Pharmacokinetics of a novel extended half-life glycoPEGylated factor IX, nonacog beta pegol (N9-GP) in previously treated adult, adolescent, and pediatric patients with hemophilia B – results from two phase 3 trials

P-W-143

A Tiede;[1] F Abdul-Karim;[2] M Carcao;[3] P Persson;[4] WHO Clausen;[4] S Kearney;[5] T Matsushita;[6] C Negrier;[7] J Oldenburg;[8] E Santagostino;[9] G Young[10]

[1]Hannover Medical School, Hannover, Germany; [2]National Blood Centre, Kuala Lumpur, Malaysia; [3]Hospital for Sick Children, Toronto, Canada; [4]Novo Nordisk A/S, Søborg, Denmark; [5]Children's Hospital and Clinics of Minnesota, MN, USA; [6]University Hospital, Nagoya, Japan; [7]Hôpital Edouard Herriot, University Claude Bernard Lyon 1, Lyon, France; [8]University Clinic Bonn, Bonn, Germany; [9]Angelo Bianchi Bonomi Hemophilia and Thrombosis Center, IRCCS Ca' Granda Foundation, Maggiore Hospital Policlinico, Milan, Italy; [10]Children's Hospital Los Angeles, University of Southern California Keck School of Medicine, Los Angeles, CA, USA

Objective

- Evaluate the pharmacokinetics (PK) of nonacog beta pegol (N9-GP) in adult/adolescent and pediatric patients with hemophilia B in two phase 3 trials (paradigm™2 and paradigm™5).

Conclusions

- Due to the long half-life of N9-GP, the mean trough value for most patients (≥17%) was in the range of mild hemophilia (factor IX [FIX] activity 5–40%)[1] after once-weekly dosing at 40 IU/kg.
- As expected, children exhibited lower plasma FIX activity levels and incremental recoveries versus adults/adolescents. All these values were higher than data from standard FIX products.
- The mono-exponential decay of FIX plasma activity profile (which differs from other long-acting products)[2] favorably supports once weekly N9-GP prophylactic dosing in patients with hemophilia B.

Introduction

- Nonacog beta pegol (N9-GP), a glycoPEGylated recombinant factor IX (FIX) product with an extended half-life, was developed for the prevention and treatment of bleeds with less frequent dosing in patients with hemophilia B.[3,4]
- In two phase 3 trials, N9-GP was well tolerated and provided effective once-weekly prophylaxis and bleed control in adult/adolescent (paradigm™2)[4] and pediatric (paradigm™5)[5] patients with hemophilia B.
- Here, we evaluate the pharmacokinetic (PK) characteristics of N9-GP across all age groups.

Methods

- N9-GP PK parameters were assessed in adults/adolescents (paradigm™2) and children (paradigm™5).
- Previously treated patients with hemophilia B (FIX activity ≤2%), no history of FIX inhibitors, and ≥50 or ≥150 exposure days to any FIX product (children aged 0–12 yrs and adults/adolescents aged ≥13 yrs, respectively) were included.
- PK data are presented for N9-GP 40 IU/kg across all age groups.
- Predose FIX activity levels were measured in all patients to assess steady-state trough levels.
- Single-dose PK were assessed across all ages, while steady-state PK were assessed in adults/adolescents.
- Blood samples for PK assessment were collected up to 168 h (1 week) after N9-GP administration.
- Standard PK parameters were estimated based on plasma FIX activities assessed with the 1-stage clotting assay and N9-GP as the reference standard.
- PK parameters were also assessed according to race and body mass index (BMI) for adults/adolescents.

An electronic version of the poster can be viewed by scanning the Quick Response (QR) code. The QR code is intended to provide scientific information for personal use only. The poster should not be altered and may not be reproduced without permission from the authors.

Results

- Thirty-four patients with full PK profiles (9 adults/adolescents and 25 children) on 40 IU/kg were included in the analysis.
- Fifty-four patients (29 adults/adolescents and 25 children) with predose FIX measurements at steady state were included in the analysis of trough levels at steady state.
- PK parameters for all patients dosed with N9-GP at 40 IU/kg are shown in Table 1.

Table 1 PK parameters for N9-GP 40 IU/kg (geometric means).

Geometric mean (CV%)	Children (single-dose PK only) 0–6 yrs[a] (n=12)	Children (single-dose PK only) 7–12 yrs[a] (n=13)	Adults/adolescents[a] Single dose (n=9)	Adults/adolescents[a] Steady state (n=9)
IR, IU/mL / IU/kg	0.015 (7.3)[b]	0.016 (16.18)	0.022 (14.5)	0.019 (21.06)
Clearance, mL/h/kg	0.8 (13.0)	0.6 (21.9)	0.4 (20.4)	0.4 (12.3)
Half-life, h	69.6 (15.8)	76.3 (25.5)	85.1 (21.8)	110.8 (11.8)
FIX activity 168 h postdose[c], IU/mL	0.084 (16.28)	0.109 (18.89)	0.16 (34.4)	0.31[d] (17.3)
$AUC_{(0–inf)}$, IU×h/mL	46.2 (14.1)	56.2 (19.1)	86.9 (22.3)	141.3 (17.4)

$AUC_{(0–inf)}$, area under the curve–from time zero to infinity; CV, confidence interval; FIX, factor IX; IR, incremental recovery at t=30 min; n, number of patients for each PK parameter; PK, pharmacokinetic. [a]Samples were not available for all patients for some PK parameters. [b]n=11. [c]FIX activity 168 h postdose; values based on n=11, 12, 8, and 8, respectively. [d]Steady-state value corresponding to predosing for next dose, i.e., trough level.

- After single-dose administration, half-life was 85.1 h in adults/adolescents versus 69.6 and 76.3 h in children aged 0–6 and 7–12 yrs, respectively (Table 1).
- Single-dose PK profiles were as expected for the different age groups (Figure 1).
- There were no apparent differences in single-dose PK parameters:
 - Across race groups for all ages (White, n=21; Black, n=1; Asian, n=9; and Others, n=3).
 - Between normal-weight (BMI 18.5–24.9 kg/m^2; n=5) and overweight (BMI ≥25 kg/m^2; n=4) individual adult/adolescent patients.

Figure 1 Single-dose PK of N9-GP 40 IU/kg in all age groups.

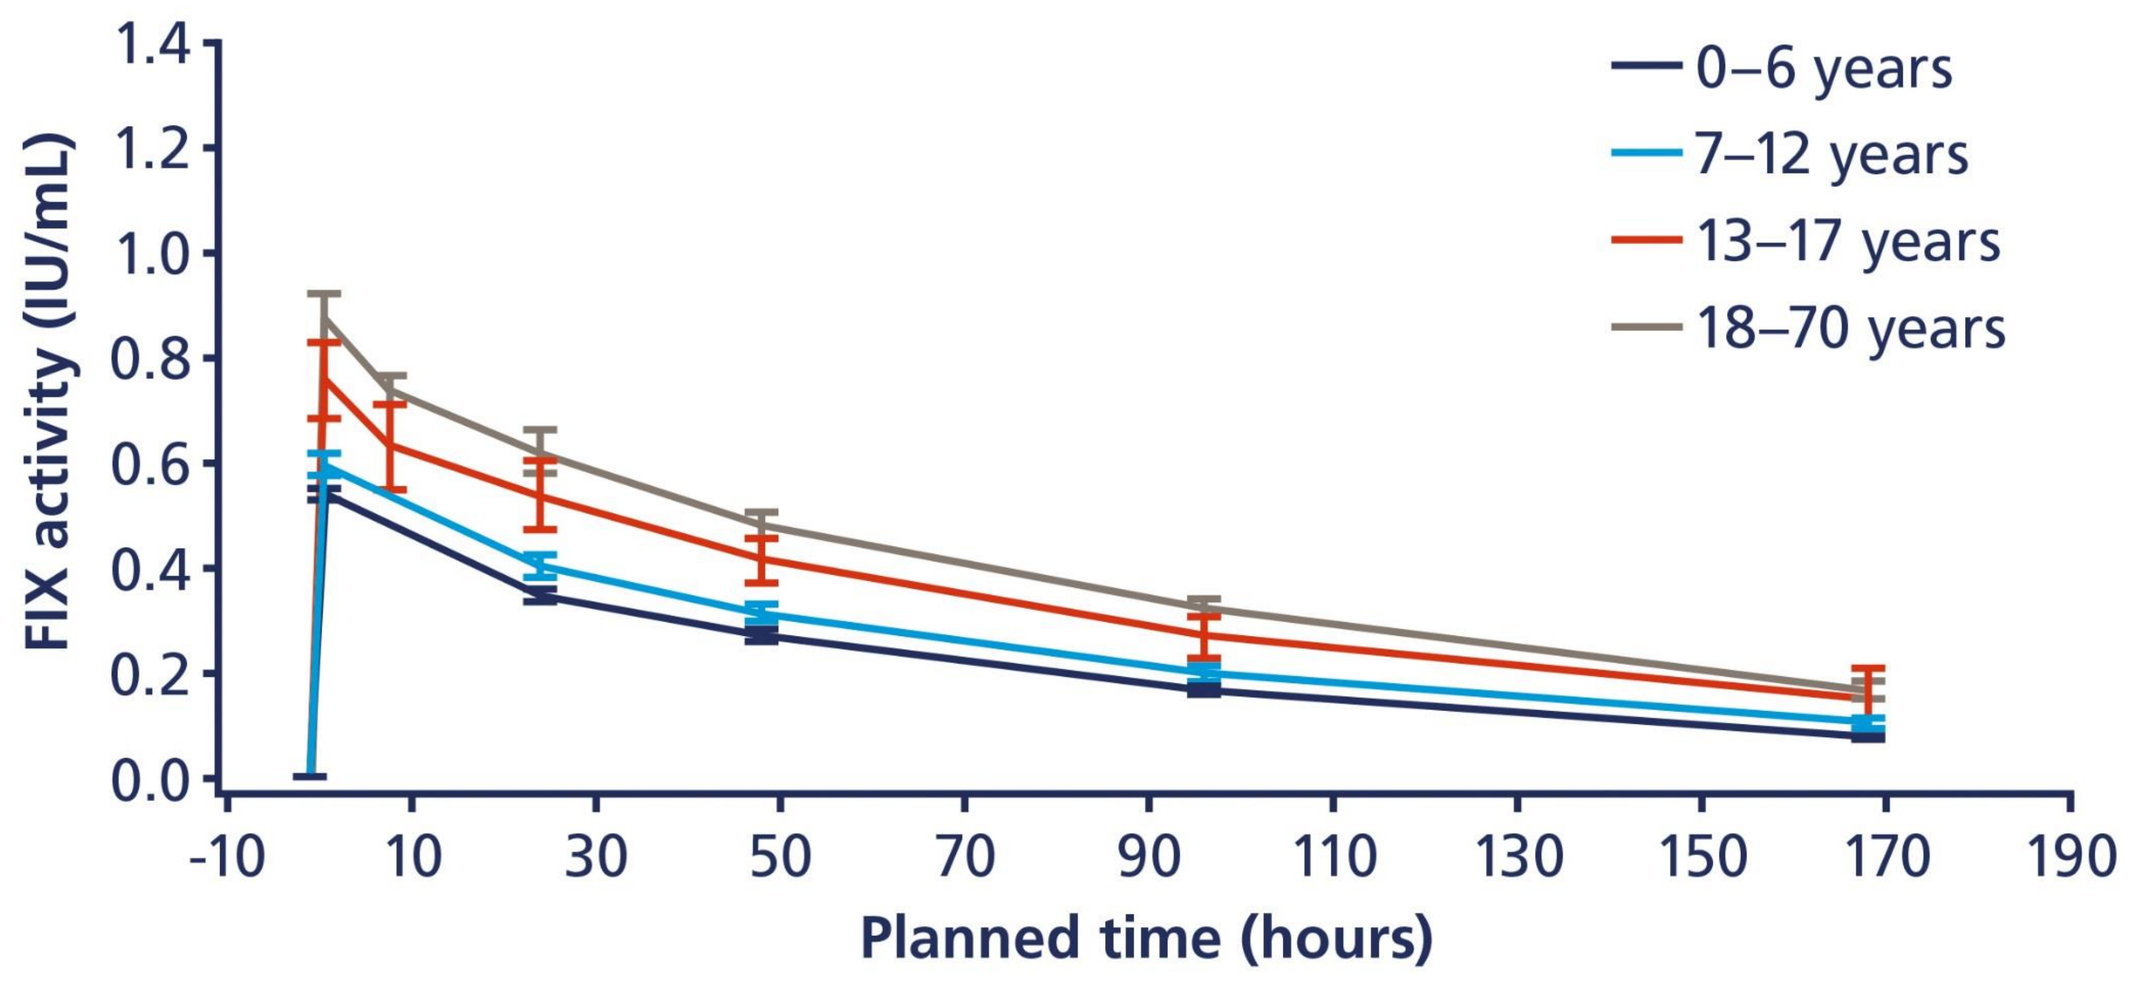

0–6 years
7–12 years
13–17 years
18–70 years
FIX activity (IU/mL)
1.4
1.2
1.0
0.8
0.6
0.4
0.2
0.0
-10
10
30
50
70
90
110
130
150
170
190
Planned time (hours)

Data show mean ± standard error. FIX, factor IX; N9-GP, nonacog beta pegol; PK, pharmacokinetic.

- Overall, plasma FIX activity levels were lower in children of all ages than in adults/adolescents.
 - Incremental recovery for single-dose PK was lower for children of all ages versus adults/adolescents, whereas clearance per kg of body weight was higher (Table 1).
- At steady state, N9-GP half-life was 110.8 h and mean trough FIX level was 0.31 IU/mL at 168 h postdose in adults/adolescents (Table 1).
 - Estimated FIX trough at steady state based on predose measurements were 0.17 and 0.27 IU/mL for ≤12 and ≥13 yrs, respectively.

Figure 2 Single-dose and steady-state PK of N9-GP 40 IU/kg in adults/adolescents.

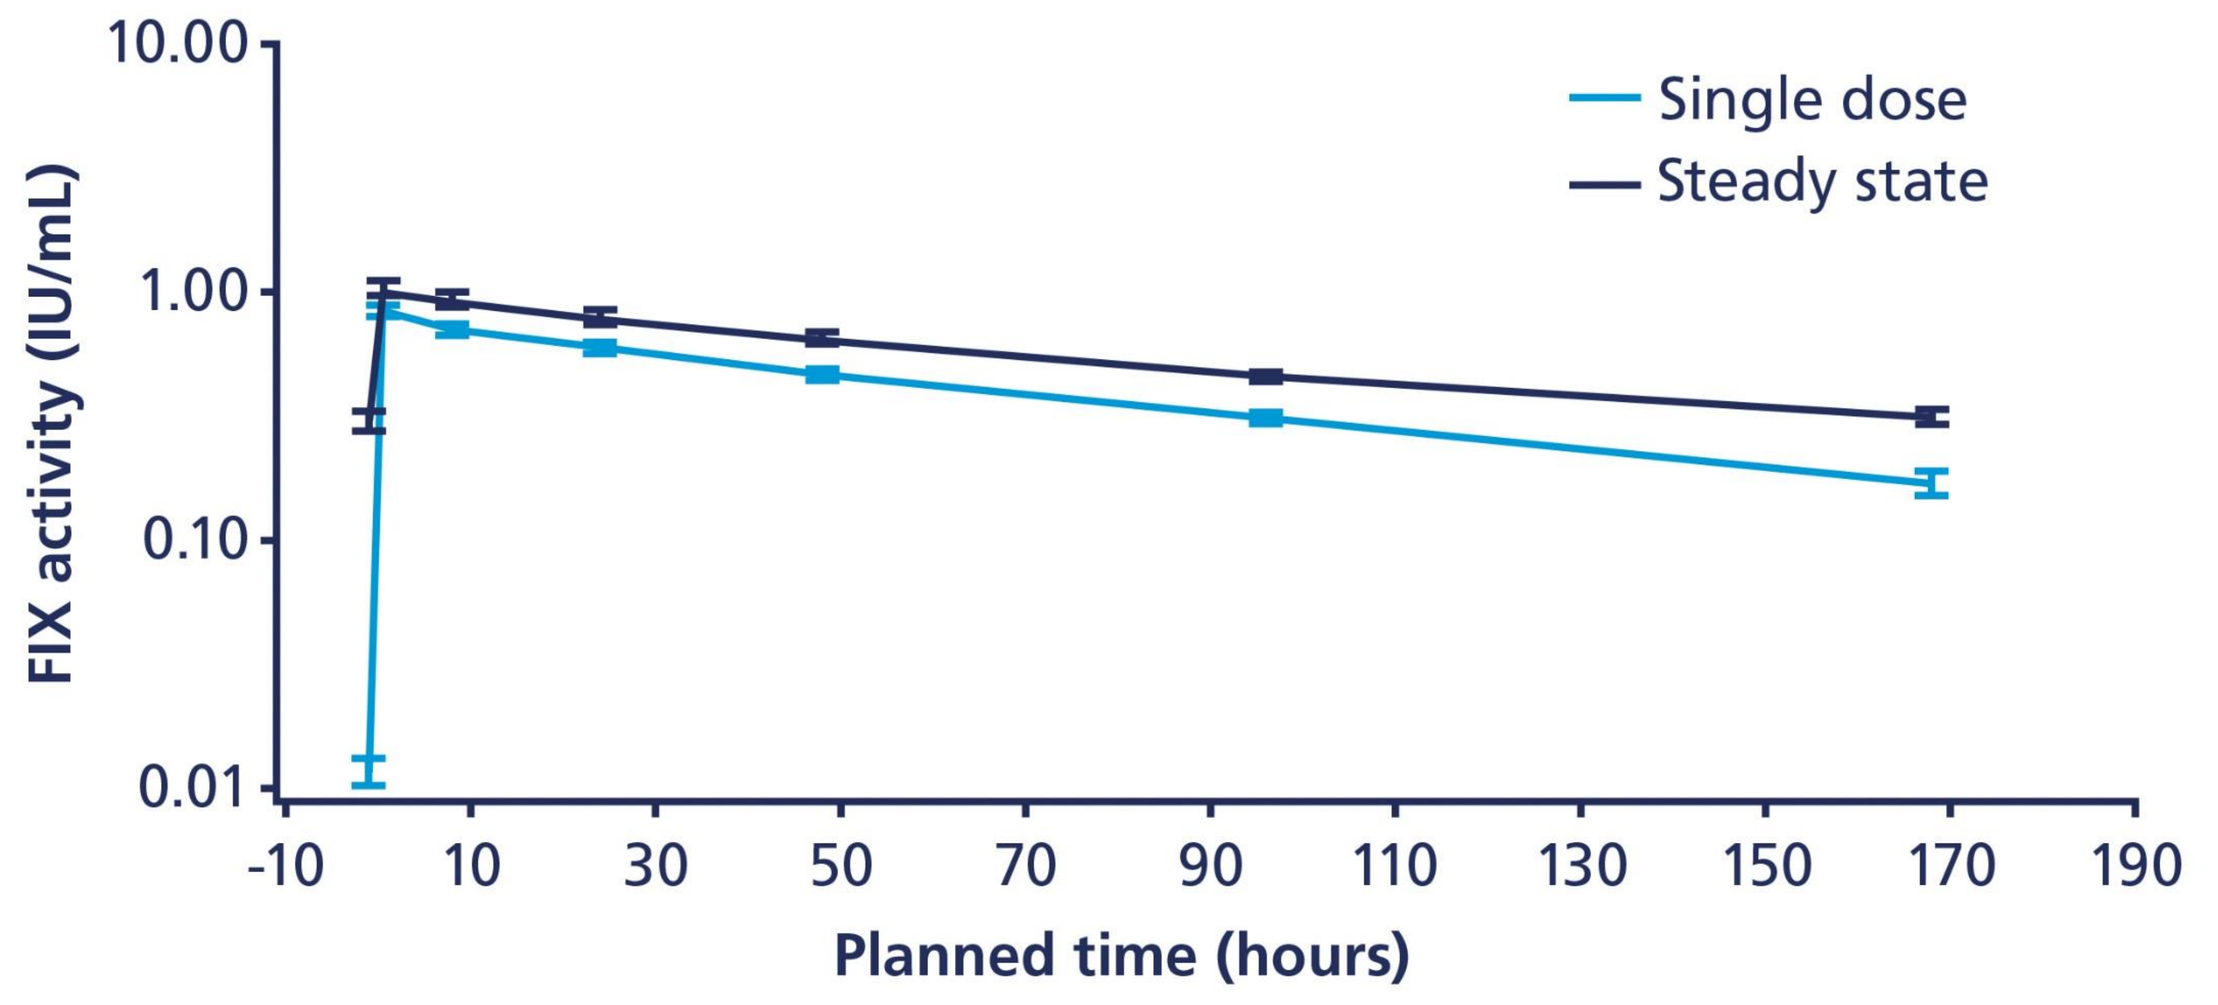

Single dose
Steady state
FIX activity (IU/mL)
10.00
1.00
0.10
0.01
-10
10
30
50
70
90
110
130
150
170
190
Planned time (hours)

Data show mean ± standard error. FIX, factor IX; N9-GP, nonacog beta pegol; PK, pharmacokinetic.

- In adults/adolescents, plasma FIX activity profiles followed a mono-exponential decay after both single-dose administration and at steady state (Figure 2).
 - At all time points, FIX activity was proportionally higher at steady state than after single-dose administration.

References

1. Srivastava A, *et al. Haemophilia* 2013;19:e1–e47.
2. Powell J, *et al*, *New Engl J Med* 2013;369:2313–2323.
3. Negrier C, *et al*. *Blood* 2011;118:2695–2701.
4. Collins PW, *et al*. *Blood* 2014;124:3880–3886.
5. Carcao M, *et al*. *Journal of Thrombosis and Haemostasis* 2016;eprint.

Conflict of interest disclosure

AT has received grants and/or personal fees for lectures and consultancy from Bayer, Baxter/Baxalta, Biogen Idec, Biotest, Boehringer Ingelheim, CSL Behring, Leo Pharma, Novo Nordisk, Octapharma, Pfizer, and Sobi. **FA-K** has no COI to declare. **MC** has received honoraria for participating in advisory boards, speaker fees, and research support from Baxalta, Bayer, Biogen, Biotest, CSL Behring, Novo Nordisk, Octapharma, and Pfizer. **PP** and **WHOC** are employees of Novo Nordisk. **SK** is an advisory board member for Biogen, and is a local principal investigator for clinical trials with Biogen and Novo Nordisk. **TM** has received speaker fees and is an advisory board member for Baxalta, Bayer, Biogen, Chugai, CSL Behring, Novo Nordisk, and Octapharma, and has received research funding from Baxalta, Bayer, CSL Behring, Kaketsuken, and Novo Nordisk. **CN** has received research support and/or is a principal investigator for Alnylam, Baxter, Bayer, Biogen Idec/Sobi, CSL Behring, Inspiration, Novo Nordisk, Octapharma, and Pfizer, has participated in scientific advisory board activities and has received honoraria and/or travel support from Baxter, Bayer, Biogen Idec/Sobi, CSL Behring, LFB, Novo Nordisk, Octapharma, and Pfizer. **JO** has received reimbursement for symposia/congress attendance, honoraria for speaking and/or consulting, and research support from Baxter, Bayer, Biogen Idec, Biotest, Chugai, CSL Behring, Grifols, Novo Nordisk, Octapharma, Pfizer, Roche, and Sobi. **ES** has an unrestricted research grant from Pfizer, and is a speaker and/or advisory board member for Bayer, Baxter/Baxalta, Biogen Idec, CSL Behring, Grifols, Kedrion, Novo Nordisk, Octapharma, Pfizer, Roche, and Sobi. **GY** is a consultant for Alnylam, has received speaker fees and is an advisory board member for Baxalta, Bayer, Biogen Idec, CSL Behring, Kedrion, Novo Nordisk, and Roche.

These trials were sponsored by Novo Nordisk and are registered with ClinicalTrials.gov (NCT01333111 and NCT01467427).
The authors acknowledge the data analysis contribution of Torben Colberg (former Novo Nordisk employee) and the medical writing assistance of Jo Fetterman (PAREXEL).
Presented at the World Federation of Hemophilia (WFH) World Congress, July 24–28, 2016, Orlando, FL, USA.

PosterSessionOnline.com

Poster Presented at: WFH 2016 World Congress Orlando, USA July 24-28

DOI: 10.3252/pso.eu.WFH2016.2016

Pharmacokinetics
Jasper Jacobsen
143--PP-W
WFH2016