Pharmacokinetics of a plasma-derived von VWF/FVIII concentrate (Voncento®) in adult/adolescent and pediatric subjects with severe hemophilia A (SWIFT-HA and SWIFTLY-HA studies)

Lissitchkov T.[1], Mamonov V.[2], Buevich E.[3], Kuliczkowski K.[4], Goranov S.[5], Kłoczko J.[6], Klukowska A.[7], Stankovic S.[8], Gercheva L.[9], Chernova T.[10], Hellmann A.[11], Dmoszyńska A.[12], Zawilska K.[13], Skotnicki A.[14], Iosava G.[15], Djambas Khayat C.[16], Romashevskaya I.[17], Aleinikova O.[18], Stasyshyn O.[19], Vashchenko L.[20], Lopez M.[21], Pompa M.[22], Rogosch T.[23], Joch C.[23], Seifert W.[23]

[1]SHAT 'Joan Pavel', Sofia, Bulgaria; [2]National Research Center for Hematology, Moscow, Russia; [3]GOUVPO Altaysky State Medical University of Roszdrav, Barnaul, Russian Federation; [4]Independent Public Clinical Hospital No. 1, Wroclaw, Poland; [5]UMHAT 'Sveti Georgi', Plovdiv, Bulgaria; [6]University Hospital, Bialystok, Poland; [7]Department of Pediatrics, Hematology and Oncology of Warsaw Medical University, Warsaw, Poland; [8]University Clinic of Hematology, Skopje, Macedonia, the Former Yugoslav Republic; [9]MHAT 'Sveta Marina', Varna, Bulgaria; [10]FGU Kirov Scientific Research Institute of Hematology and Blood Transfusion, Kirov, Russian Federation; [11]Academic Clinical Centre - Hospital, Medical University, Gdansk, Poland; [12]Independent Public Clinical Hospital No. 1, Lublin, Poland; [13]Dept. of Hematology. J.Strus Hospital, Poznan, Poland; [14]Independent Public Health Care University Hospital, Department of Clinical Hematology, Krakow, Poland; [15]Joint Stock Hematology and Transfusiology Research Institute, Tbilisi, Georgia; [16]Hospital Hôtel Dieu de France, Beirut, Lebanon; [17]Republican Research Centre of Radiation Medicine and Human Ecology, Gomel; [18]Republican Scientific and Practical Centre of Children Oncology and Heamatology, Minsk, Belarus; [19]Institute of Blood Pathology and Transfusion Medicine, Lviv, Ukraine; [20]Dnipopetrovsk Kafedra Pediatric Hospital, Dnipopetrovsk, Ukraine; [21]Guatemala City Hospital Roosevelt, Guatemala, Guatemala; [22]Monterrey Nuevo Leon OCA Hospital (MIRC), Monterrey Nuevo Leon, Mexico; [23]Clinical Development, CSL Behring, Marburg, Germany

Introduction and Objective

Voncento® is a plasma-derived, high-concentration, low-volume, high-purity concentrate, which contains a high level of high-molecular-weight multimers and a VWF:FVIII ratio of ~2.4:1. The SWIFT (“**S**tudies with von **Wi**llebrand factor/**F**ac**t**or VIII”) program is evaluating this product in hemophilia A and VWD patients in accordance with the European clinical and pediatric guidelines. Within this program the pharmacokinetics (PK) of pretreated children <12 yrs, adolescents and adult patients with severe hemophilia A (FVIII:C <1%) was studied.

Study Design

Subjects with severe hemophilia A received Voncento® as a single bolus infusion of 50 IU FVIII/kg body weight (n=31: children < 12 years of age; n=16: adults/adolescents). PK parameters for VWF and FVIII were derived from plasma concentration values collected prior to dosing and at 0.5, 4, 8, 24, and 48 h after infusion. PK parameters comprised Incremental recovery (IR), Half-life ($t_{1/2}$), AUC, C_{max}, Mean residence time (MRT), Clearance (CL), Volume of distribution at steady state (V_{ss}).

PK parameters of FVIII:C

Mean (SD) concentration profiles [IU/mL] of FVIII:C

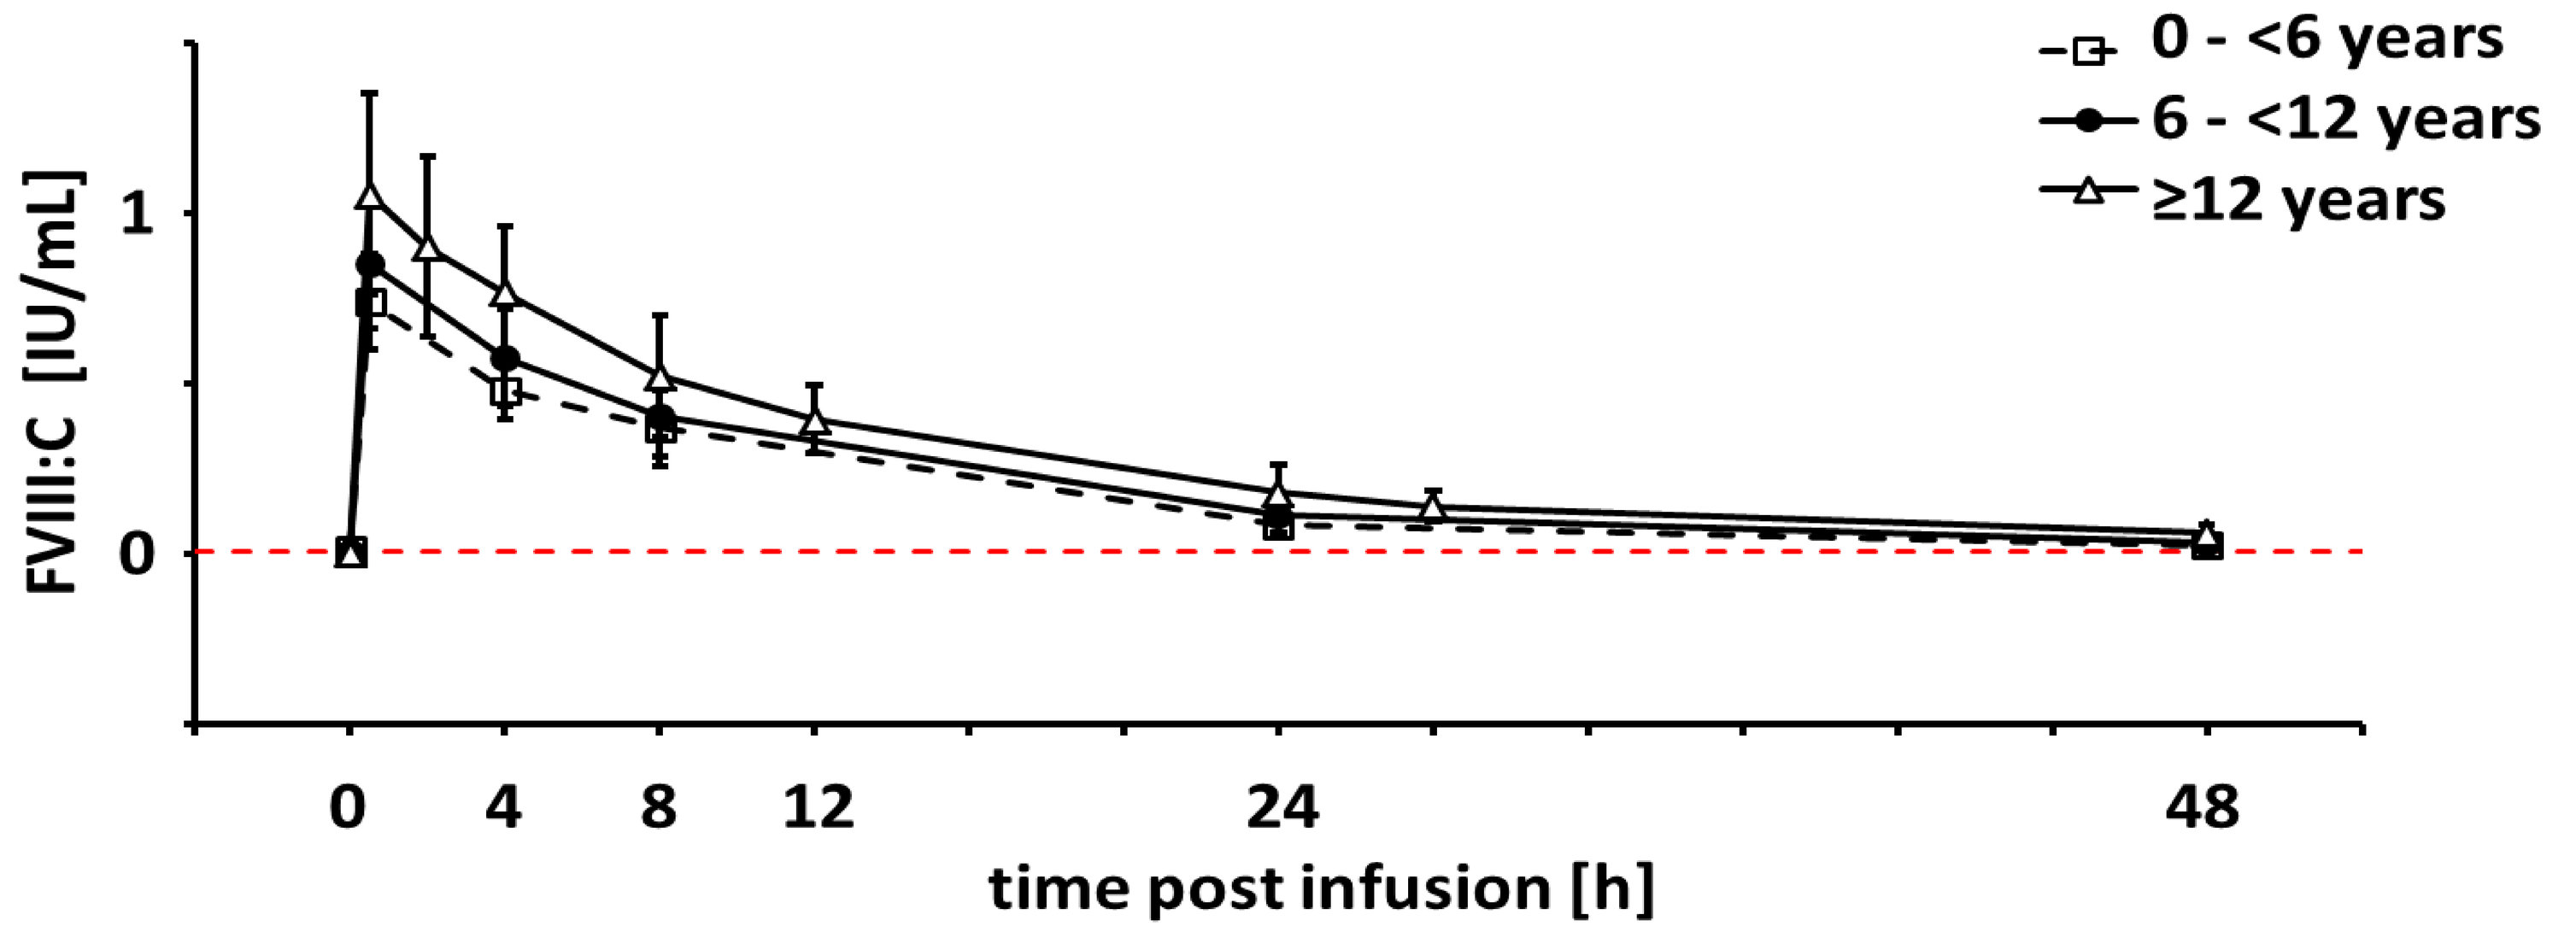
0 - <6 years
6 - <12 years
≥12 years
FVIII:C [IU/mL]
1
0
0
4
8
12
24
48
time post infusion [h]

FVIII:C	Age 0 - <6 years		Age 6 – 12 years		Age ≥ 12 years	
	N	Median (range)	N	median (range)	N	median (range)
IR [(IU/mL)/(IU/kg)]	15	0.015 (0.009-0.019)	16	0.016 (0.010-0.026)	16	0.021 (0.011-0.032)
$t_{1/2}$ [h]	15	9.62 (7.75-18.20)	16	10.00 (8.89-12.50)	16	13.74 (8.78-18.51)
MRT [h]	15	13.51 (7.95-17.38)	16	13.89 (12.11-17.07)	16	16.62 (11.29-26.31)
AUC_{0-48} [h*IU/mL]	15	8.23 (3.96-11.04)	16	9.90 (6.16-17.62)	16	13.09 (7.04-21.79)
C_{max} [IU/mL]	15	0.75 (0.46-0.94)	16	0.84 (0.51-1.21)	16	1.07 (0.57-1.57)
CL [mL/(h*kg)]	15	6.22 (4.22-11.34)	16	4.88 (2.54-7.74)	16	3.82 (2.30-7.11)
V_{ss} [mL/kg]	15	75.3 (63.8-197.2)	16	71.9 (42.1-109.3)	16	61.2 (35.1-113.1)

AUC = area under the curve; Cmax = maximum plasma concentration; IU = International Unit; MRT = mean residence time; N = number of subjects;; Vss = volume of distribution at steady state; FVIII:C = Factor VIII: Coagulant

Results

Concentration-time curves showed similar profiles for the adult/adolescents PK population and subjects aged < 12 years. The results for PK parameters for Factor VIII in pediatric population (< 12 yrs) are within the range of those in patients ≥ 12 years old. As expected the median IR of FVIII in the pediatric population (0-6/6-12 yrs) was slightly lower (0.015/0.016 [IU/mL]/[IU/kg]) compared to adult/adolescents hemophilia A subjects (0.021 [IU/mL]/[IU/kg]), the median $t_{1/2}$ was shorter (9.62/10.00 h vs. 13.74 h) and the median CL was higher (6.02/4.88 vs. 3.82 mL/h/kg).

Conclusion

The results for PK parameters for Factor VIII in pediatric population (< 12 yrs) are within the range of those in patients ≥ 12 years old, thus considered as comparable with the adolescents and adults. The small differences are not considered being clinically relevant. A contemporary comprehensive development program evaluating high concentration, low volume Voncento® across all age groups in hemophilia A is now available.

Biotherapies for Life™ CSL Behring

PosterSessionOnline.com
Poster Presented at: WFH 2016 World Congress
DOI: 10.3252/pso.eu.WFH2016.2016
Pharmacokinetics
Wilfried Seifert
141--PP-W
WFH2016