USC University of Southern California

COMPARISON OF CLINICAL CHARACTERISTICS AND HEALTH CARE UTILIZATION AMONG INDIVIDUALS WITH HEMOPHILIA A AND B IN THE HEMOPHILIA UTILIZATION GROUP STUDIES (HUGS) COHORTS

Mimi Lou[1], Megan Ullman[2], Judith Baker[3], Marion Koerper[4], Joanne Wu[1], Brenda Riske[5], Randall Curtis[6], Michael B. Nichol[1]; HUGS V Group

[1]University of Southern California, CA, [2]University of Texas Health Science Center at Houston, TX, [3]University of California Los Angeles and The Center for Comprehensive Care & Diagnosis of Inherited Blood Disorders, CA, [4]University of California San Francisco, CA, [5]University of Colorado at Denver, CO, [6]Factor VIII Computing, CA

INTRODUCTION

- Numerous studies in the last decade suggest greater clinical differences between hemophilia A (HA) and hemophilia B (HB) than previously appreciated.
- Bleeding episodes in persons with HB may be less severe, occur with less frequency, result in less arthropathy and require fewer orthopedic surgeries than in hemophilia HA.[1-4]
- National U.S. and Canadian surveillance reports demonstrate a lower use of prophylaxis among HB than HA.[5,6]
- Findings have been inconclusive, due to lack of epidemiologic evidence and the low incidence of HB.

OBJECTIVE

- To evaluate clinical and treatment differences among persons with HB and HA in the United States, using two multi-state cohort studies, the Hemophilia Utilization Group Studies Parts Va (HUGS Va) and Vb (HUGS Vb).

METHODS

- In 2005-2007 and 2009-2012, persons with factor VIII or IX deficiency were enrolled in HUGS Va (HA) and HUGS Vb (HB) from six and ten federally supported Hemophilia Treatment Centers (HTCs) respectively.
- The HTCs provide comprehensive care to patients in eleven states (California, Colorado, Indiana, Massachusetts, Michigan, Mississippi, Montana, Ohio, Texas, Washington and Wyoming).
- Of 477 individuals recruited in two studies, data from 355 individuals with at least 3 participant follow-up surveys and completed follow-up clinician chart and dispensing records were included in the analyses.
- In both studies, participants or parents of pediatric enrollees completed regularly scheduled surveys, supplying information on health care utilization, treatment regimen, joint pain and motion limitation, socio-demographic and clinical characteristics.
- Twenty-four months of follow-up data were collected for bleeding episodes, outpatient procedures and visits, hospitalizations, emergency room (ER) visits and factor utilization.
- Self-reported joint pain was measured by a question that assessed pain on a five-point scale. Self-reported limitation in Joint Range of Motion (JROM) was measured by a question that assessed limitation of motion on a four-point scale.
- Annualized bleed rate (ABR) was calculated from two-year participant-reported bleeding episodes and defined as number of bleeding episodes per year.
- The comparisons between two study cohorts, HA vs. HB, were assessed using Chi-square or Fisher's Exact tests for categorical variables, and Wilcoxon-Mann-Whitney tests for continuous variables.

REFERENCES

1. Tagariello G, Iorio A, Santagostino E, Morfina M, Bisson R, Innocenti M *et al*. Comparison of the rates of joint arthroplasty in patients with severe factor VIII and IX deficiency: an index of different clinical severity of the 2 coagulation disorders. *Blood* 2009; **114**: 779-783.
2. Nagel K, Walker I, Decker K, Chan AKC, Pai MK. Comparing bleed frequency and foactor concentrate us between haemophilia A and B patients. *Haemophilia* 2011; **17**: 872-874.
3. Schulman S, Eelde A, Holmstrom M, Stahlberg G, Odeberg J, Blombacks M. Validation of a composite score for clinical severity of hemophilia. *J. Thromb Haemost* 2008; **6**: 1113-21.
4. Melchiorre D, Linari S, Romano E, Sofi F, Matucci-Cerinic M, Carulli C. Clinical, instrumental, serological and histological findings suggest that hemophilia B may be less severe than hemophilia A. *Haematologica* 2016; **101**: 219-225.
5. Centers for Disease Control and Prevention. *Summary report of UDC activity: national treatment/clinical characteristics (hemophilia)* Dec. 31, 2011.
6. Biss TT, Chan AK, Blanchette VS, Iwenofu LN, McLimont, Carcao MD. The use of prophylaxis in 2663 children and adults wit haemophilia: results of the 2006 Canadian national haemophilia prophylaxis survey. *Haemophilia* 2008; **14**: 923-30.

Table 1: Sociodemographic and Clinical Characteristics

Characteristics	Hemophilia A (N=243)	Hemophilia B (N=112)	Total (N=355)	P Value*
Age (Mean±SD)	21.3±15.2	22.1±17.6	21.6±16.0	0.91
Adults (≥18 years old)	33.2±12.4	37.9±15.0	34.7±13.3	0.09
Children (2-17 years old)	9.4±4.5	9.5±3.8	9.4±4.3	0.76
Adults (≥18 years old), N (%)	121 (49.8%)	50 (44.6%)	171 (48.2%)	0.37
Race/Ethnicity, N (%)				0.42
White/non-Hispanic	170 (70.0%)	70 (62.5%)	240 (67.6%)	
Black/non-Hispanic	10 (4.1%)	8 (7.1%)	18 (5.1%)	
Hispanic	32 (13.2%)	18 (16.1%)	50 (14.1%)	
Asian/Pacific Islander	15 (6.2%)	5 (4.5%)	20 (5.6%)	
Others ‡	16 (6.6%)	11 (9.8%)	27 (7.6%)	
Employment, N (%) £				0.16
Full-time	105 (43.2%)	53 (47.3%)	158 (44.5%)	
Part-time	54 (22.2%)	15 (13.4%)	69 (19.4%)	
Unemployed/Retired	84 (34.6%)	43 (38.4%)	127 (35.8%)	
Household Income, N (%) £				0.82
≤ $20,000	41 (18.6%)	19 (18.3%)	60 (18.5%)	
$20,001 to $39,999	51 (23.1%)	29 (27.9%)	80 (24.6%)	
$40,000 to $74,999	54 (24.4%)	24 (23.1%)	78 (24.0%)	
≥ $75,000	75 (33.9%)	32 (30.8%)	107 (32.9%)	
Hemophilia Severity, N (%)				<0.01
Severe	169 (69.5%)	55 (49.1%)	224 (63.1%)	
Moderate/Mild	74 (30.5%)	57 (50.9%)	131 (36.9%)	
Using Prophylaxis, N (%)	110 (45.3%)	34 (30.4%)	144 (40.6%)	<0.01
Adults using Prophylaxis	39 (32.2%)	12 (24.0%)	51 (29.8%)	0.28
Children using Prophylaxis	71 (58.2%)	22 (35.5%)	93 (50.5%)	<0.01
HIV/AIDS	35 (14.4%)	4 (3.6%)	39 (11.0%)	<0.01
HCV	92 (37.9%)	23 (20.5%)	115 (32.4%)	<0.01

Abbreviations: SD – standard deviation, HIV/AIDS – human immunodeficiency virus/acquired immunodeficiency syndrome, HCV – hepatitis C virus A
* P-values were calculated from Chi-square (or Fisher Exact) tests for categorical variables or Wilcoxon-Mann-Whitney tests for continuous variables;
£ Data do not add up to N=355 because of missing data;
‡ *Others include: American Indian, Alaskan Native and others*

Figure 3a & 3b: Mean Annualized Bleed Rate (ABR) by Prophylaxis and On-Demand

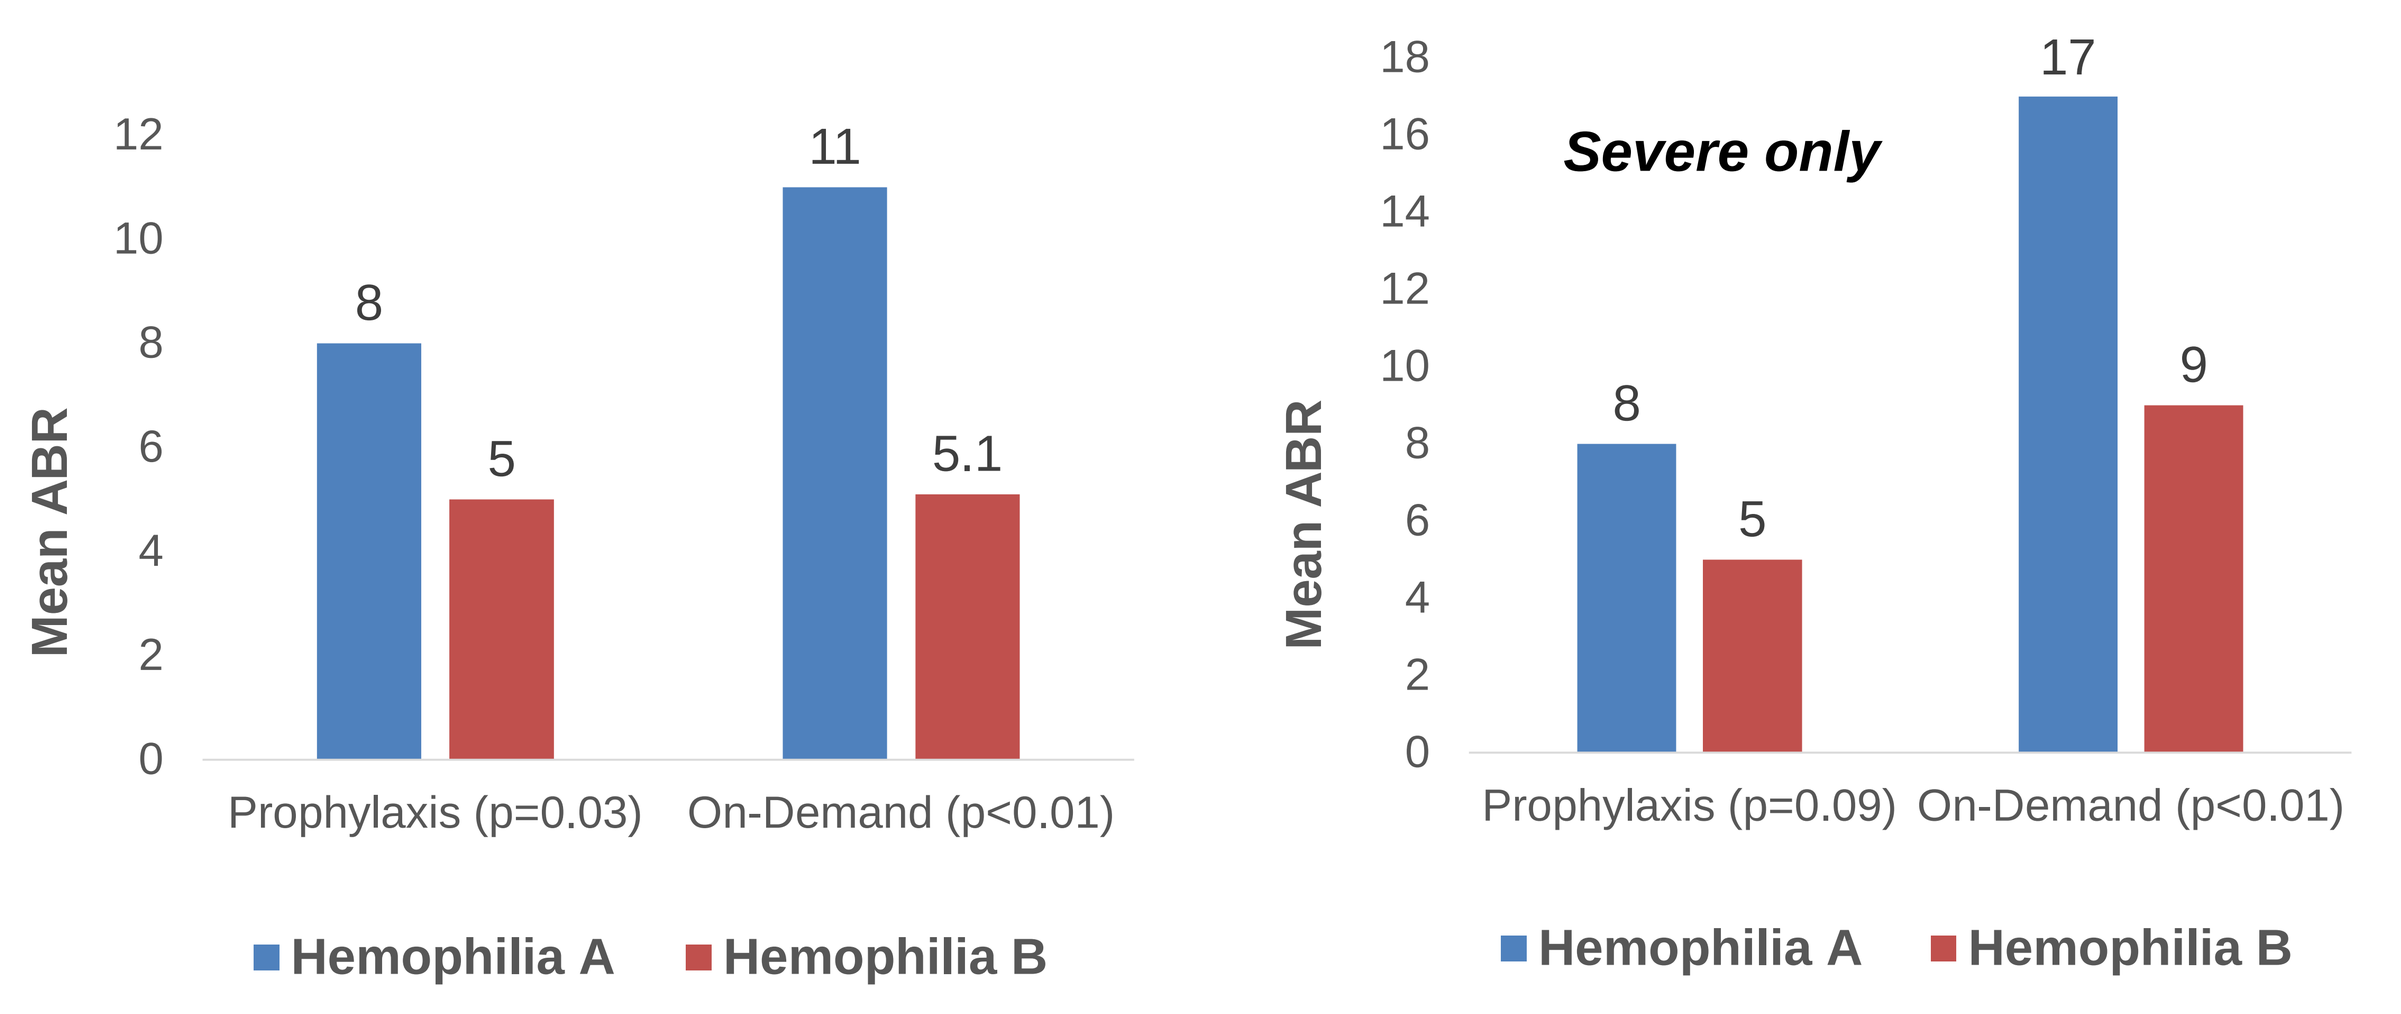
Mean ABR
12
10
8
6
4
2
0
8
5
11
5.1
Prophylaxis (p=0.03)
On-Demand (p<0.01)
Hemophilia A
Hemophilia B
Severe only
18
16
14
12
10
8
6
4
2
0
8
5
17
9
Prophylaxis (p=0.09)
On-Demand (p<0.01)
Hemophilia A
Hemophilia B

Figure 4: % Severe Patients on Prophylaxis

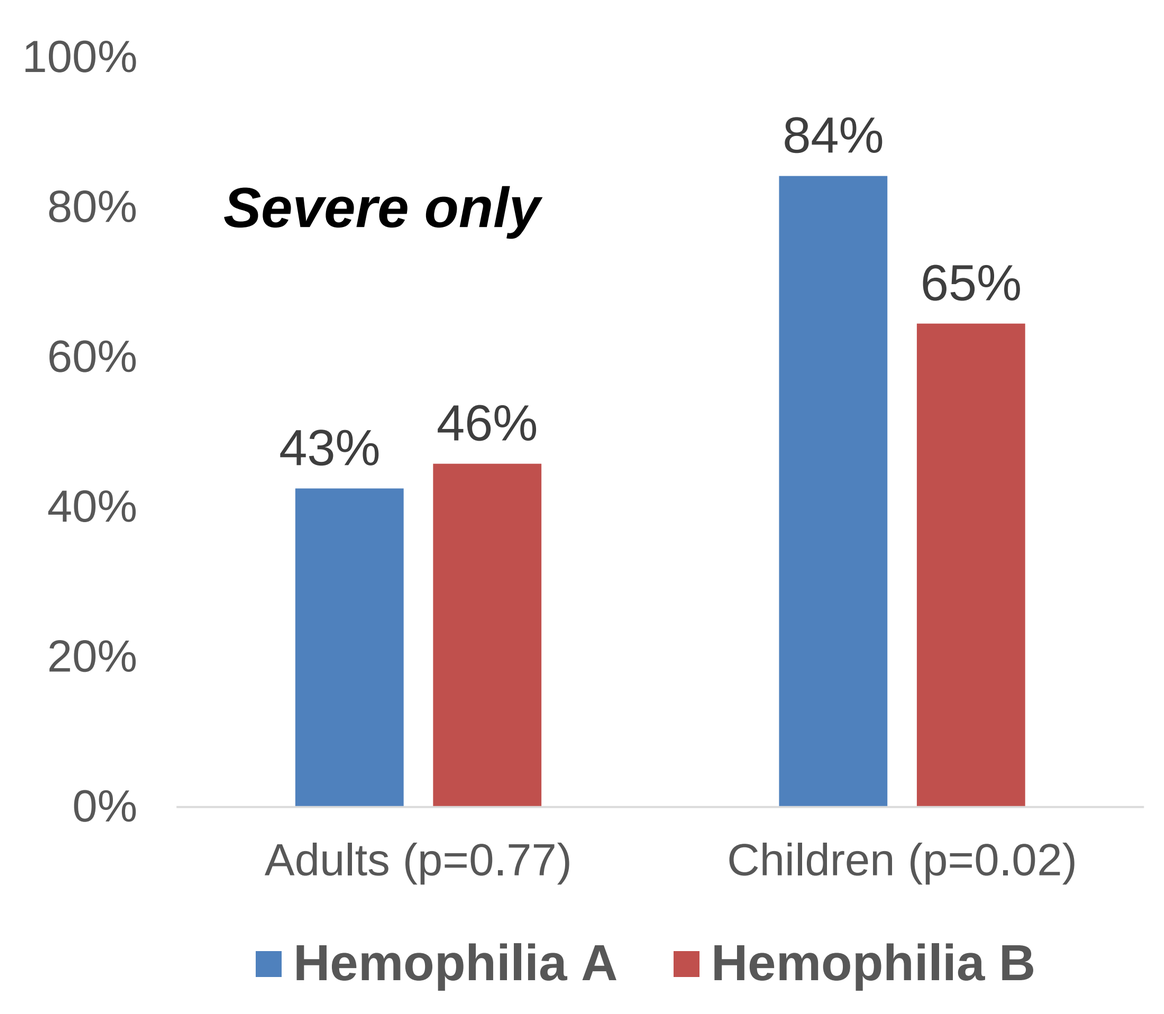
Severe only
100%
80%
60%
40%
20%
0%
43%
46%
84%
65%
Adults (p=0.77)
Children (p=0.02)
Hemophilia A
Hemophilia B

Figure 5: Mean ABR Adults vs. Children*

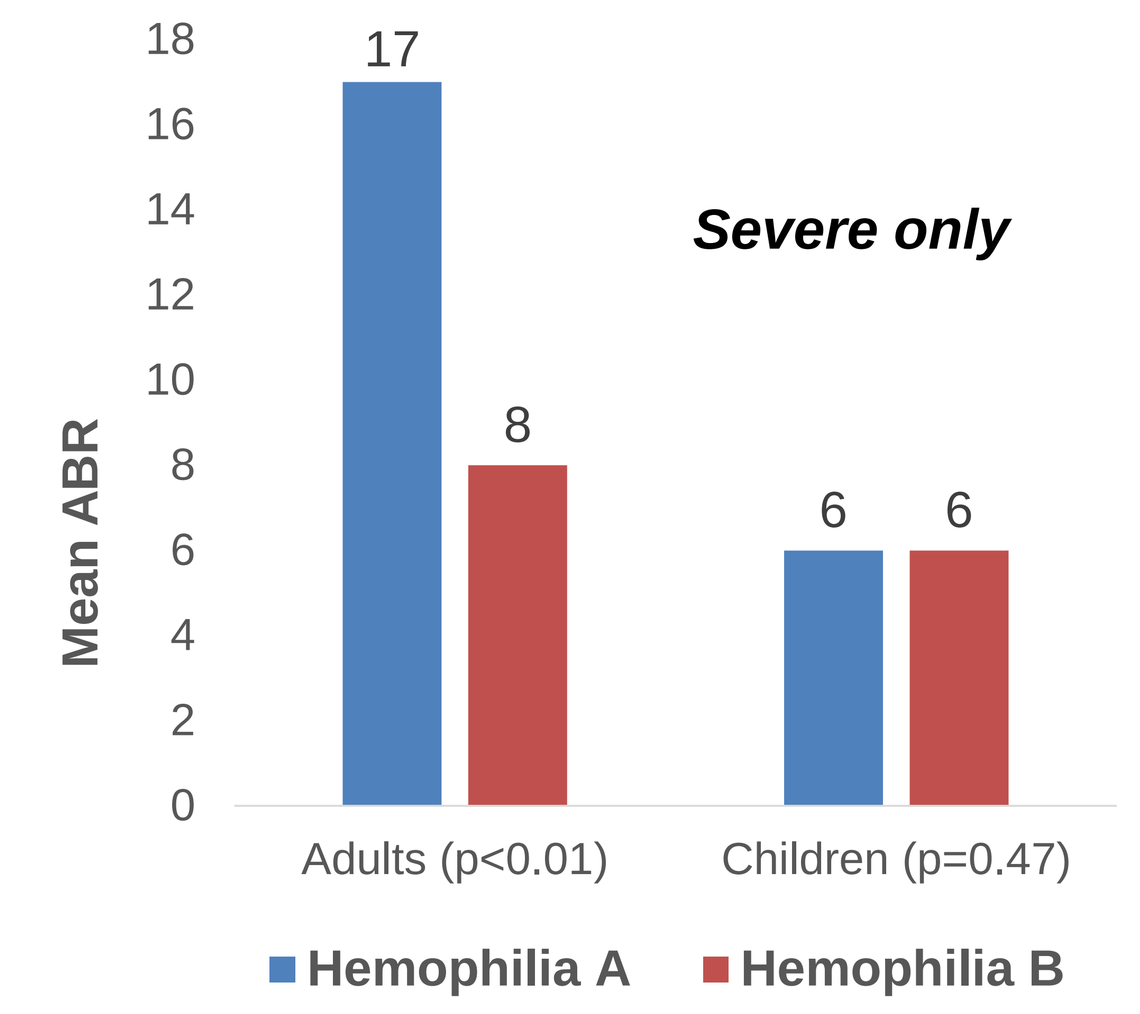
Mean ABR
18
16
14
12
10
8
6
4
2
0
17
8
6
6
Severe only
Adults (p<0.01)
Children (p=0.47)
Hemophilia A
Hemophilia B

* Including both prophylaxis and on-demand

Figure 1a: Self-reported Joint Pain (Adults)

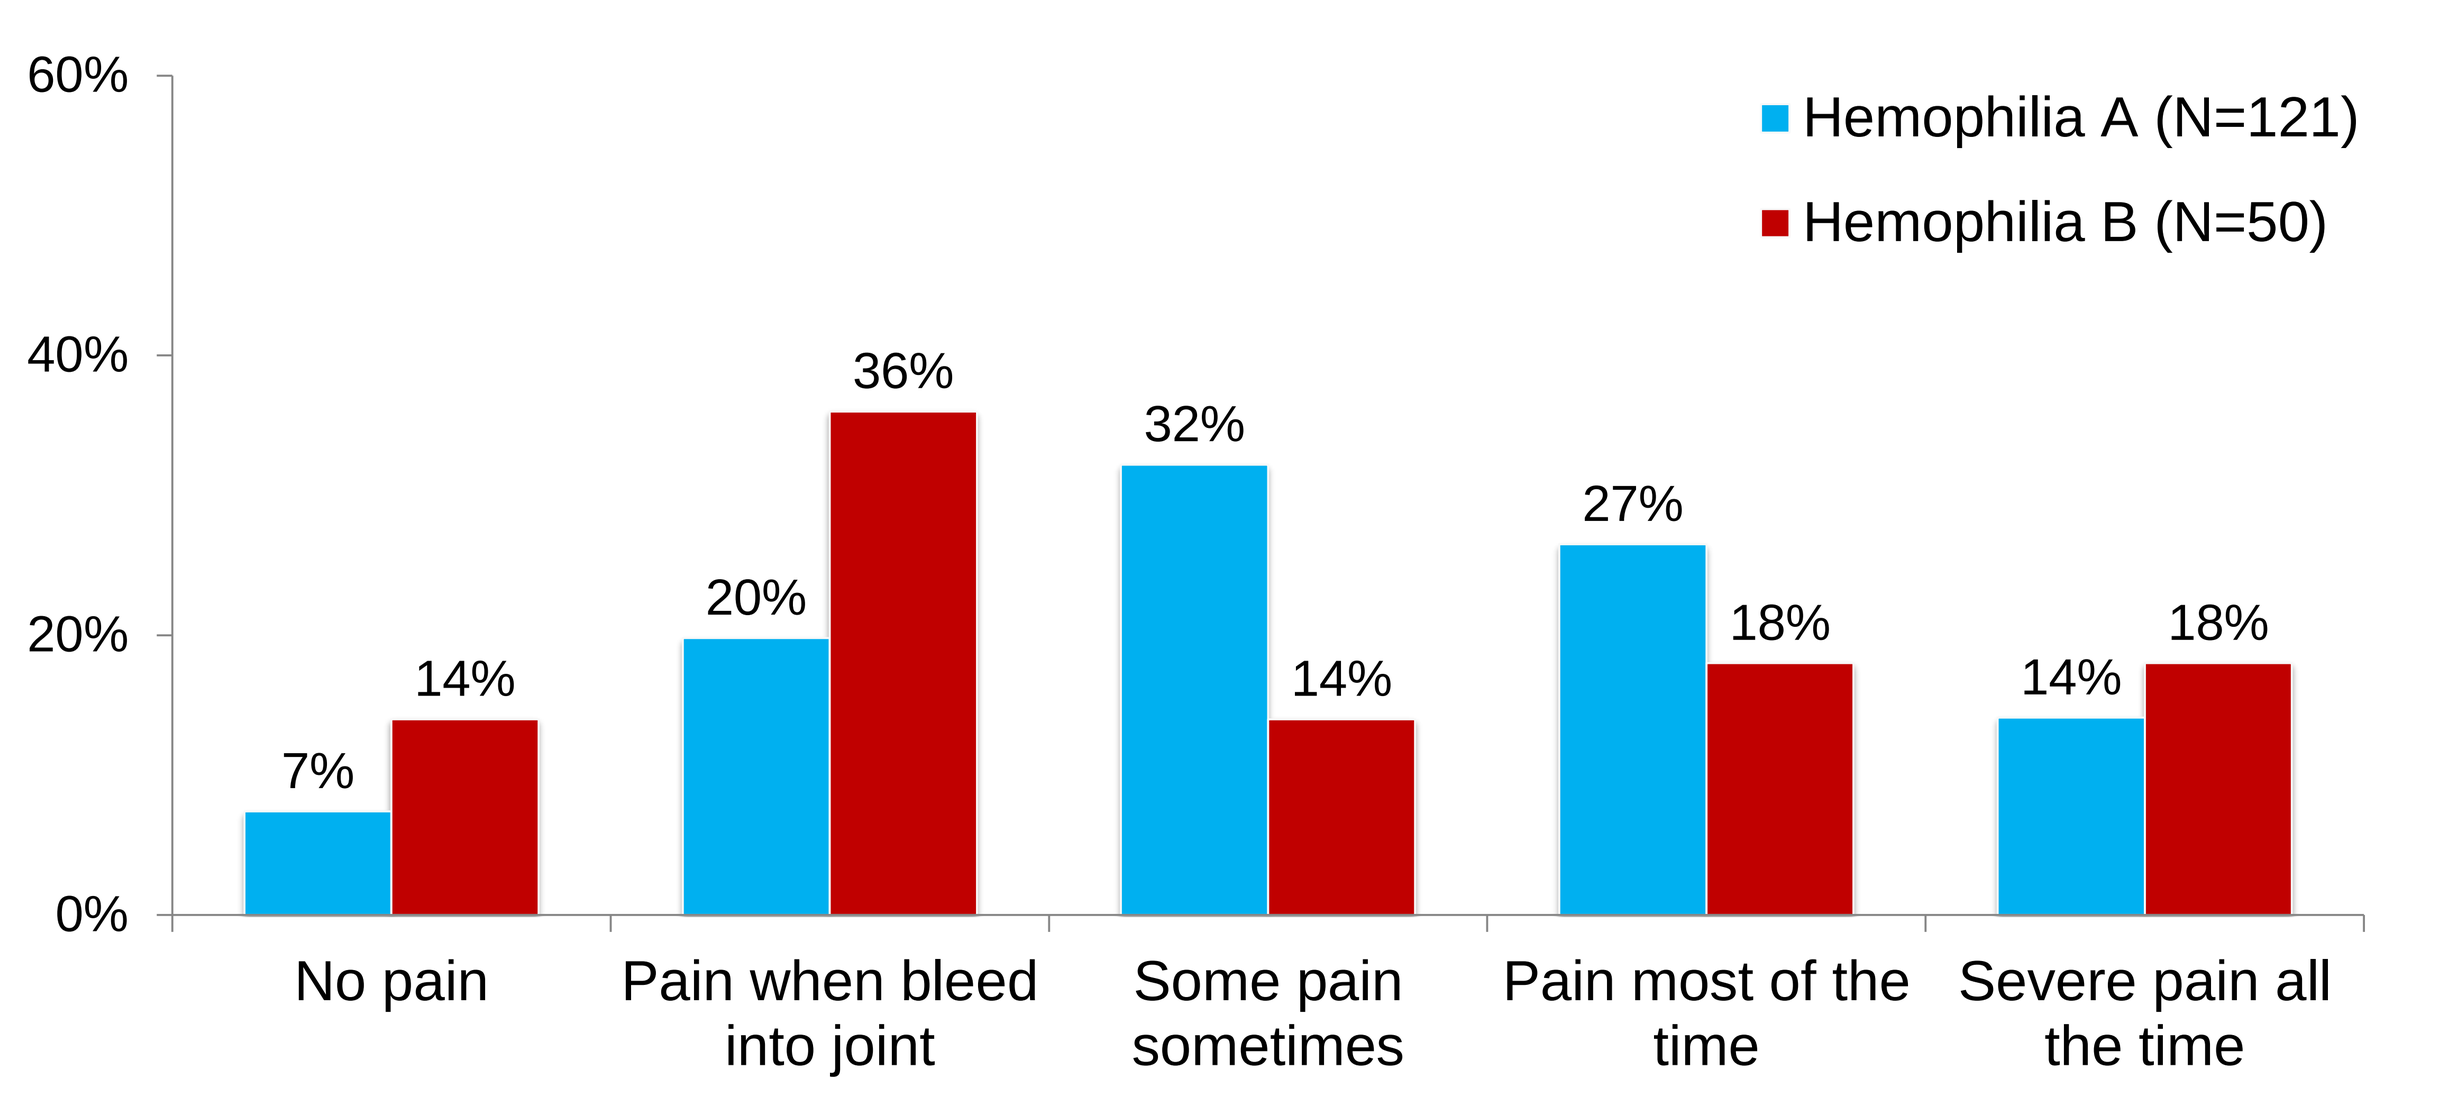
Hemophilia A (N=121)
Hemophilia B (N=50)
60%
40%
20%
0%
7%
14%
20%
36%
32%
14%
27%
18%
14%
18%
No pain
Pain when bleed into joint
Some pain sometimes
Pain most of the time
Severe pain all the time

Figure 1b: Self-reported Joint Pain (Children)

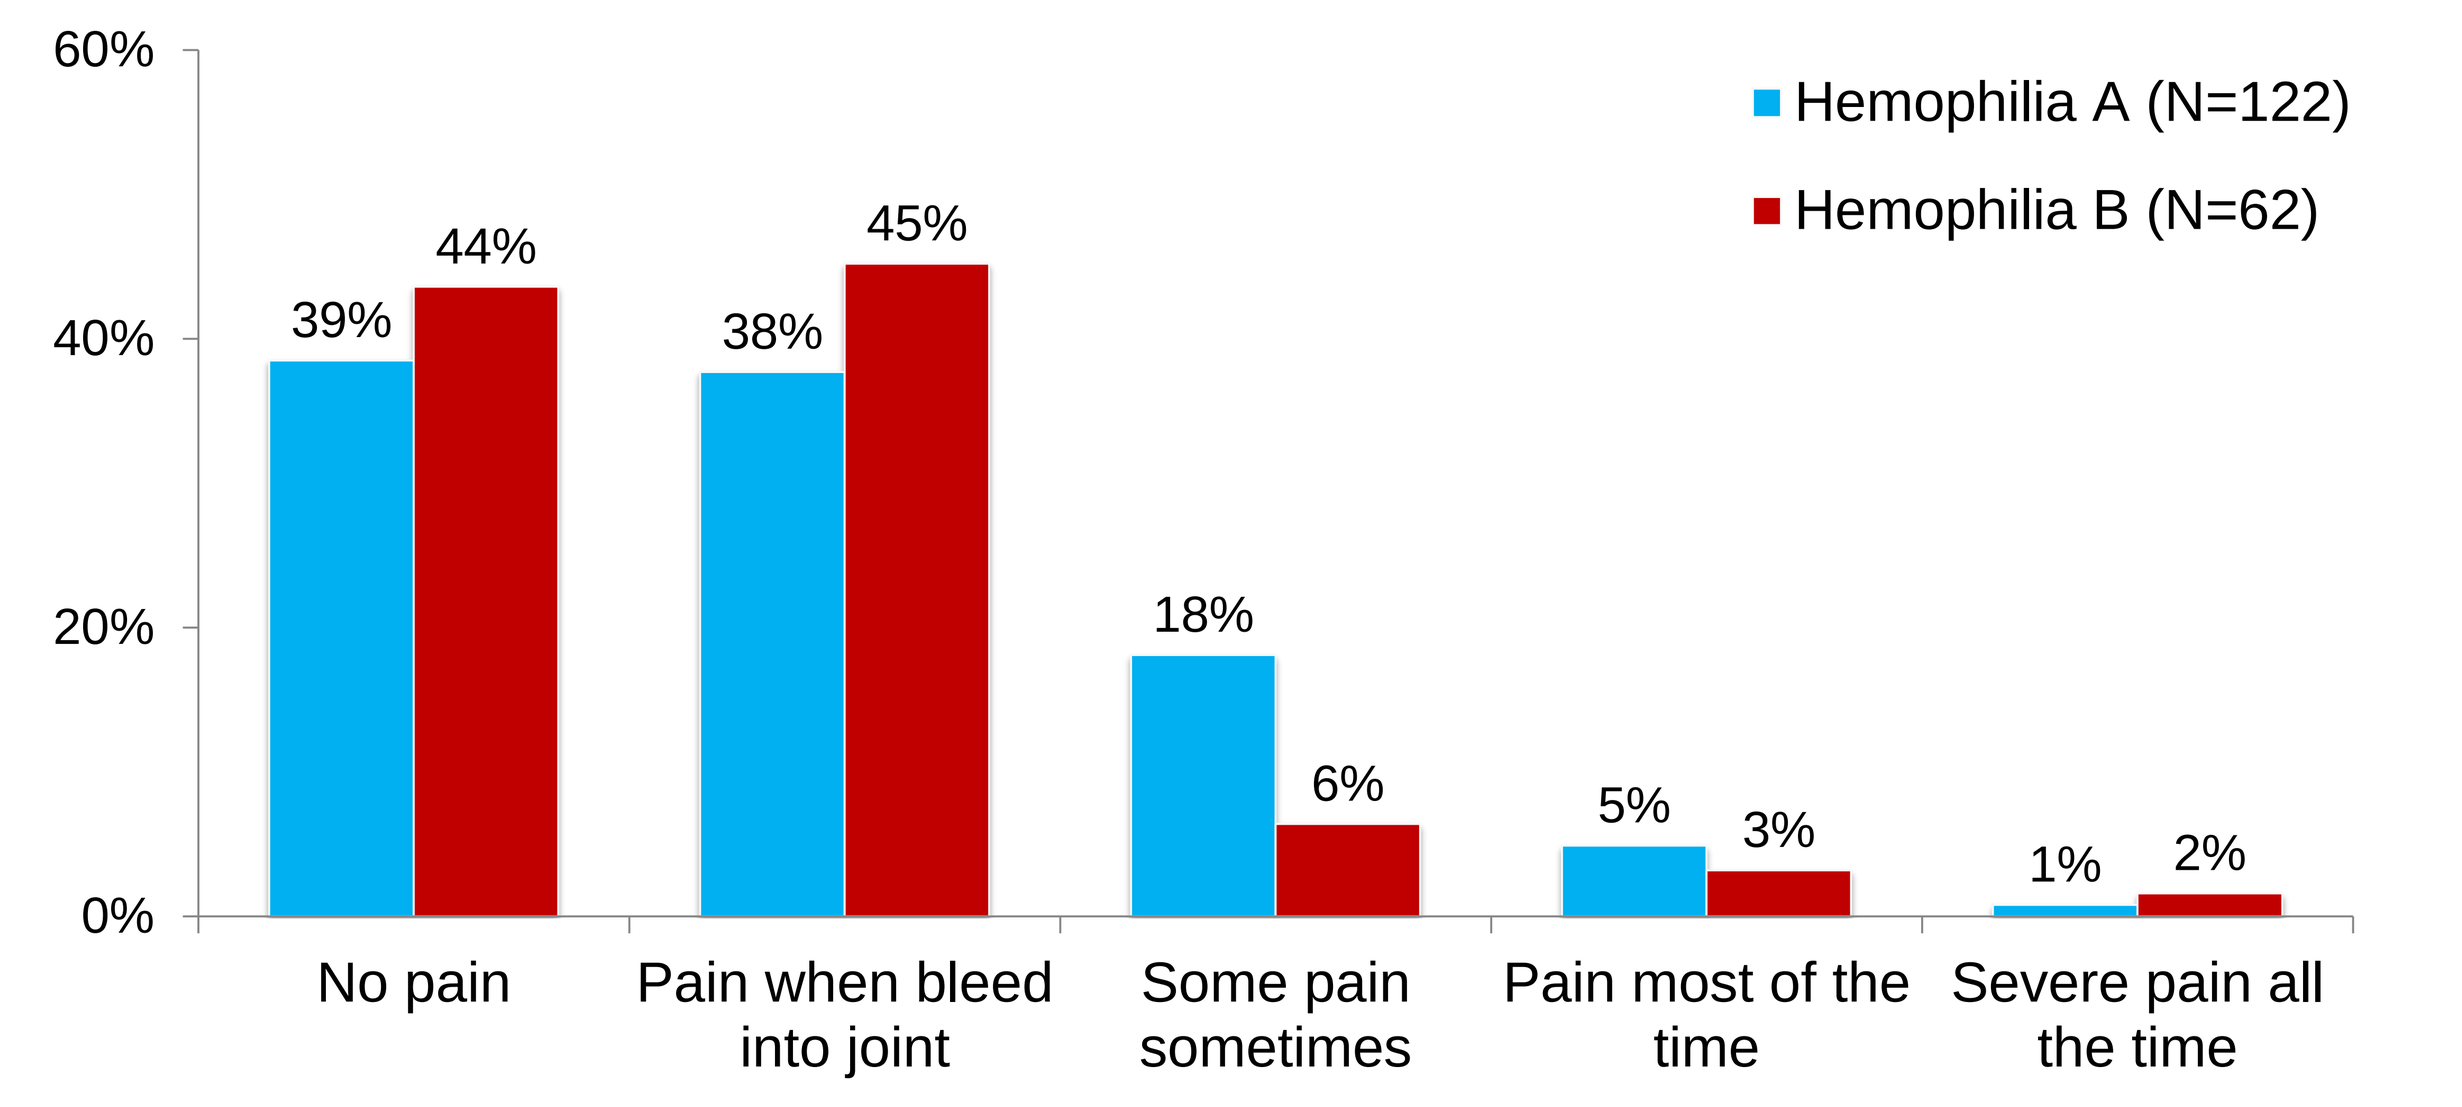
Hemophilia A (N=122)
Hemophilia B (N=62)
60%
40%
20%
0%
39%
44%
38%
45%
18%
6%
5%
3%
1%
2%
No pain
Pain when bleed into joint
Some pain sometimes
Pain most of the time
Severe pain all the time

Figure 2a: Self-reported Limitation in JROM (Adults)

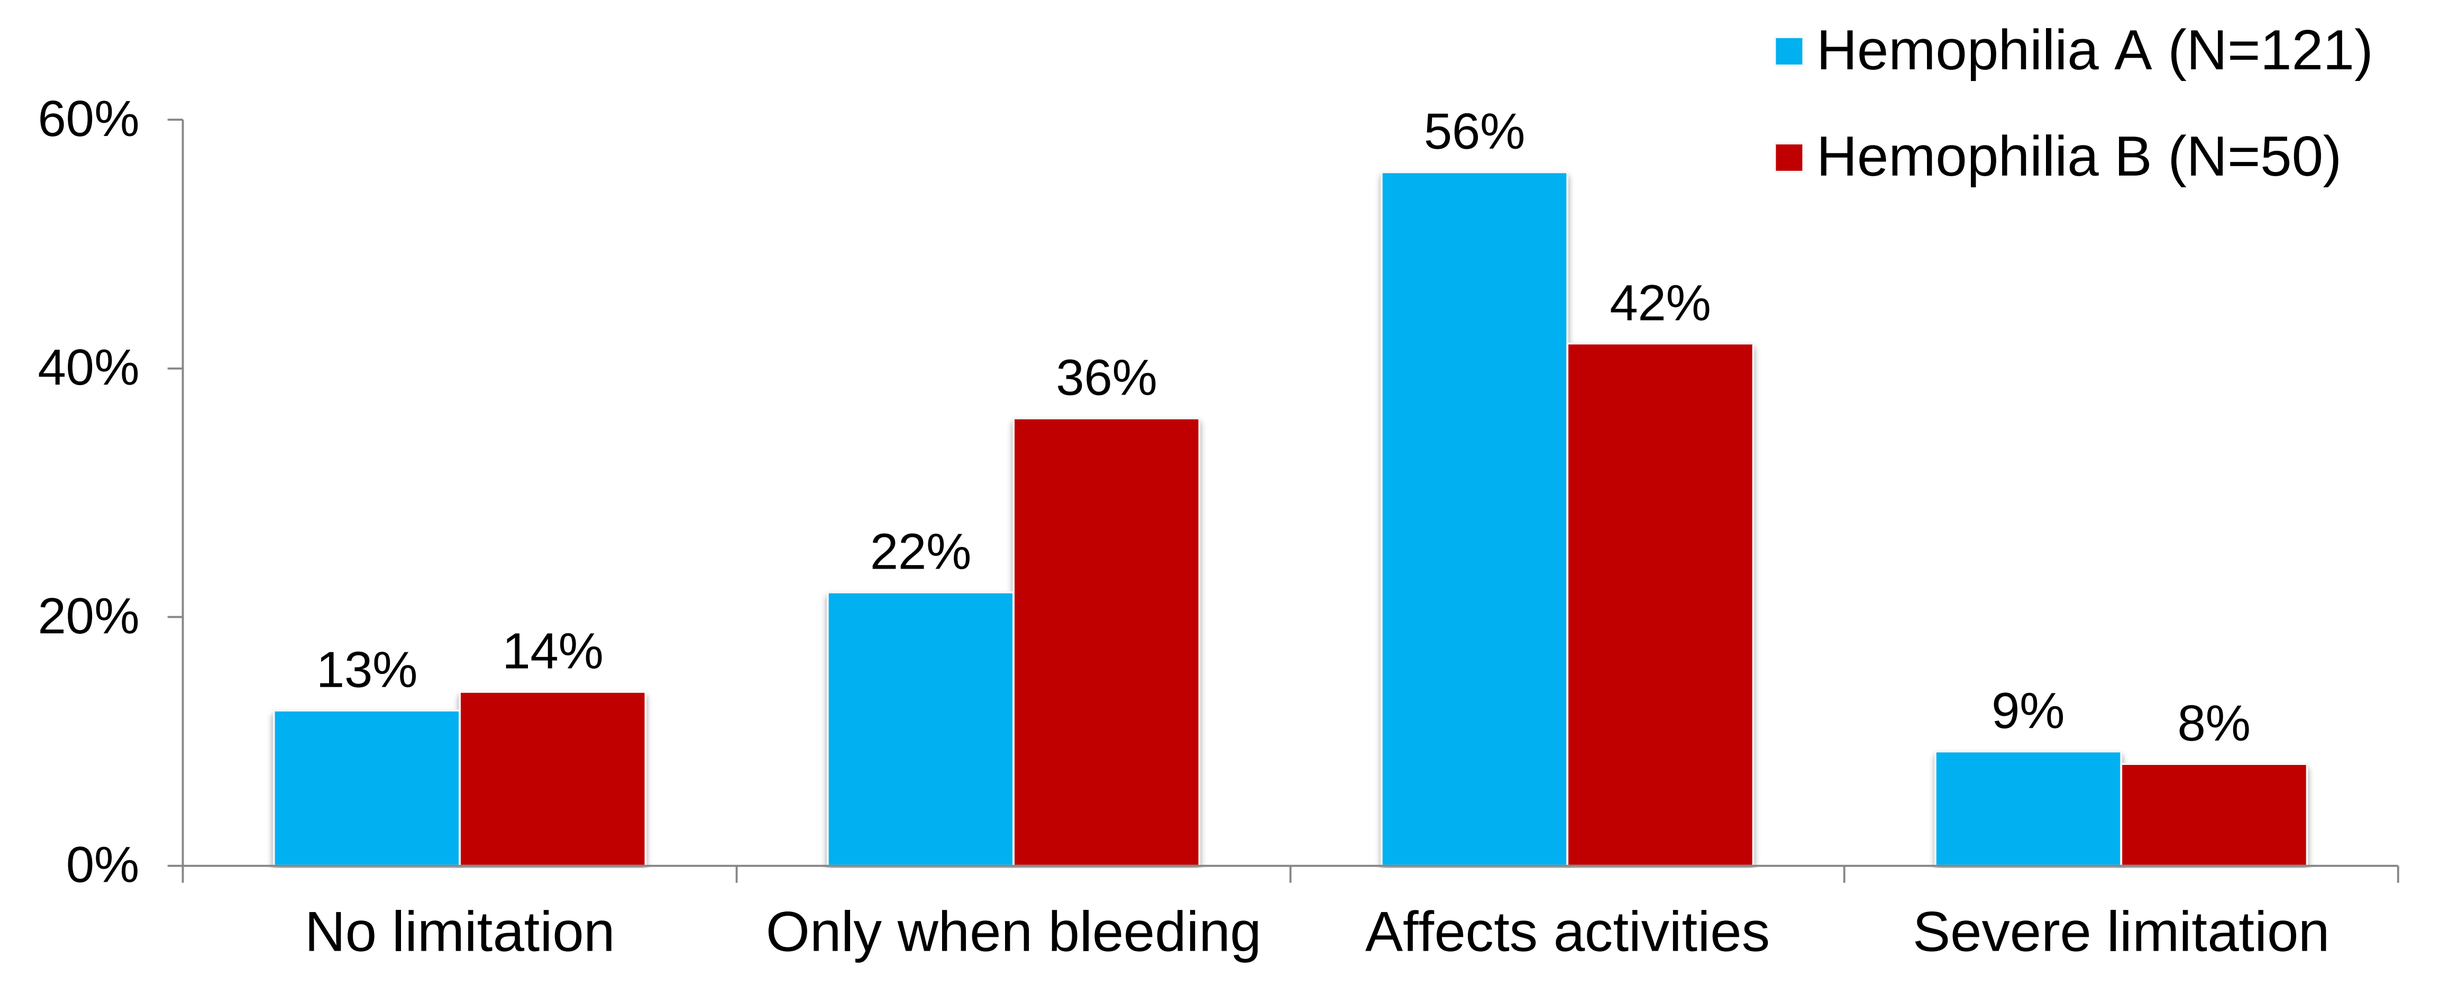
Hemophilia A (N=121)
Hemophilia B (N=50)
60%
40%
20%
0%
13%
14%
22%
36%
56%
42%
9%
8%
No limitation
Only when bleeding
Affects activities
Severe limitation

Figure 2b: Self-reported Limitation in JROM (Children)

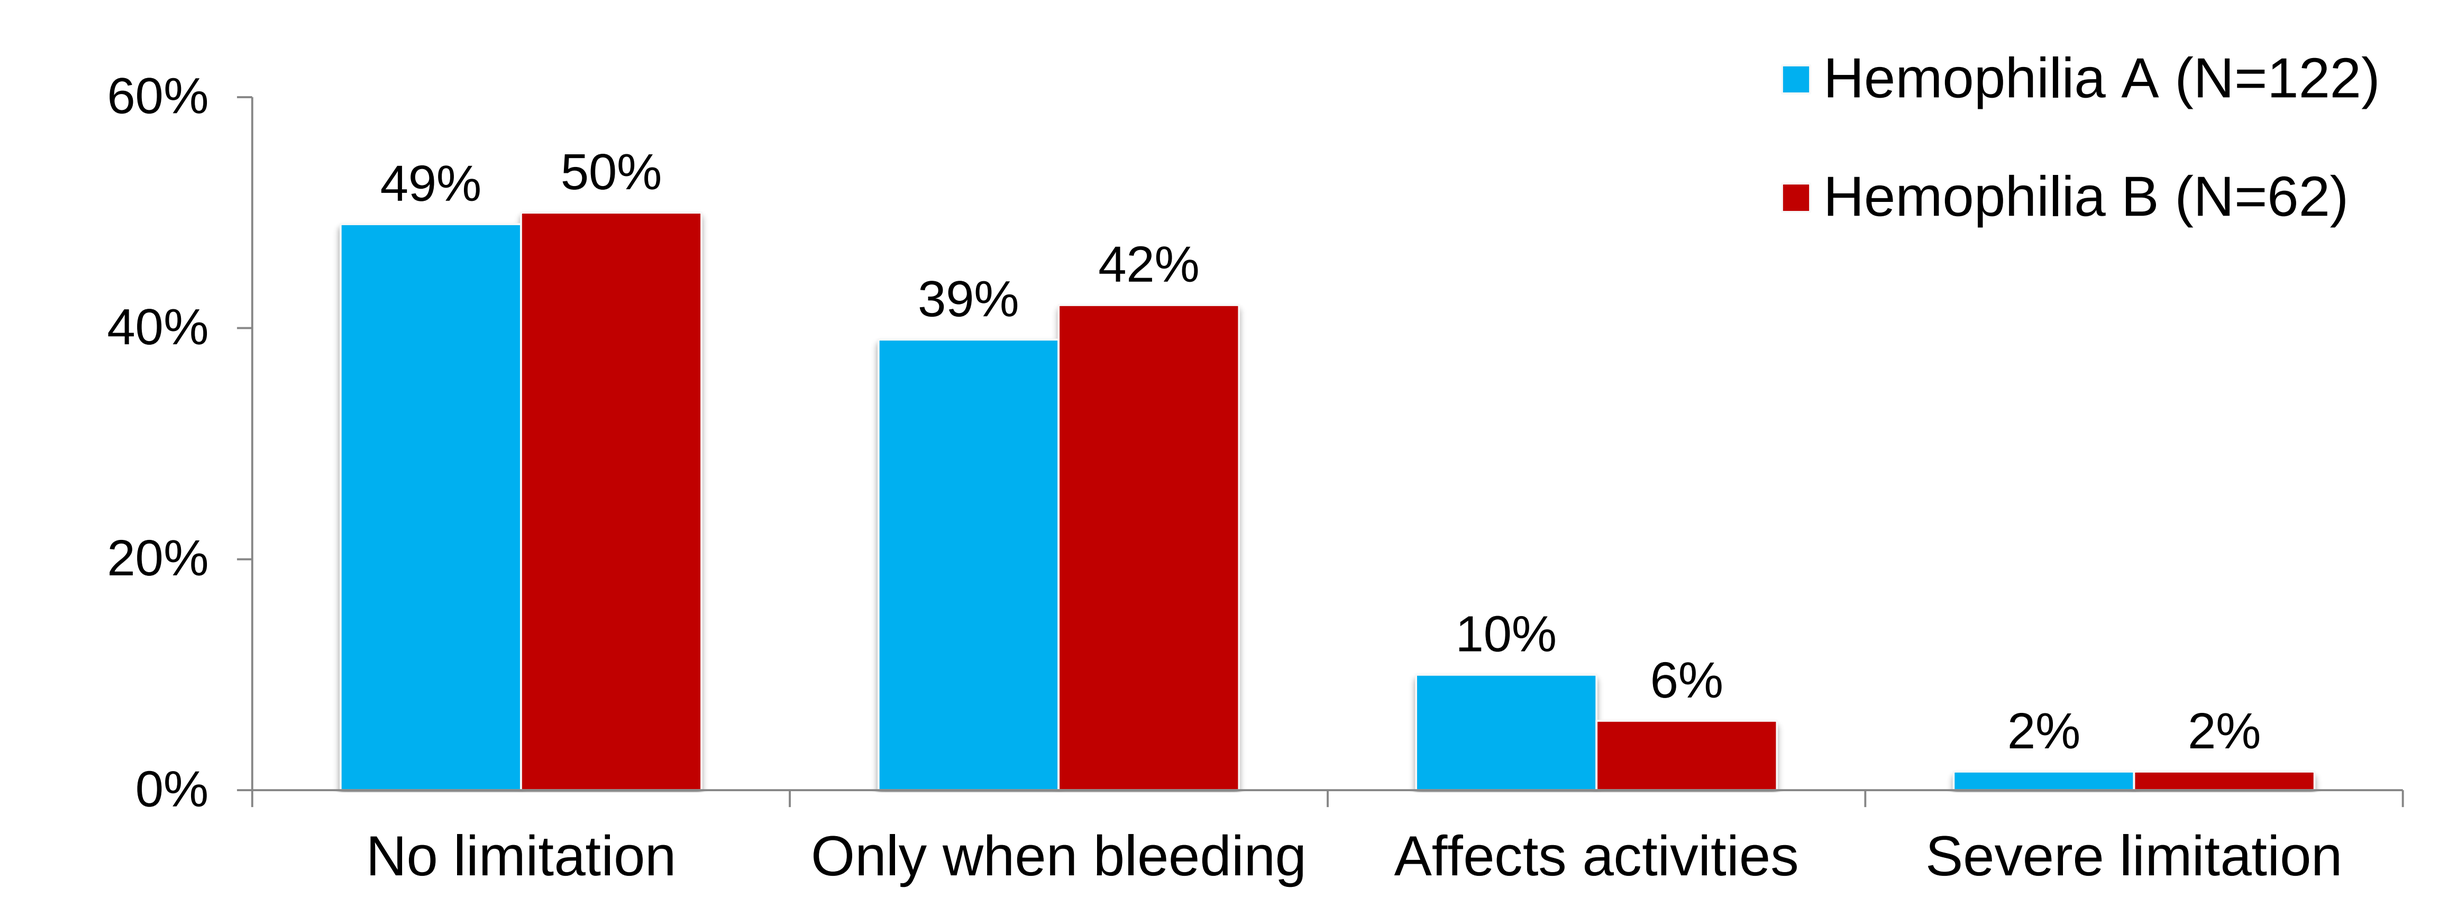
Hemophilia A (N=122)
Hemophilia B (N=62)
60%
40%
20%
0%
49%
50%
39%
42%
10%
6%
2%
2%
No limitation
Only when bleeding
Affects activities
Severe limitation

RESULTS

- Of 355 individuals in the analyses, 68% had HA and 52% were children. Both groups displayed similar socio-demographic characteristics (Table 1). Seventy percent with HA and 49% with HB had severe hemophilia.
- Self-reported joint pain was significantly different among the two groups (p=0.02): twice as many adults with HB (all severities) reported no pain compared to adults with HA (Figure 1a & 1b).
- Motion limitation affecting activities was higher in severe HA adults (65%) compared to HB adults (46%).
- Regardless of age group, severity and treatment regimen, persons with HA had significantly higher mean ABR than HB counterparts (9±10.5 vs. 5±6.5, p<0.01)
- The mean ABR among all ages and severities treating episodically was nearly two times higher in HA than HB (p<0.01) (Figure 3a).
- Among children with severe disease, a significantly (p=0.02) larger proportion with HA (84%) infused prophylactically versus HB (65%). There was no significant difference between the adults cohort (Figure 4).
- Adults with severe HA had significantly higher mean ABR than HB counterparts, regardless of treatment regimen (p<0.01). Children had similar mean ABR between HA and HB (Figure 5).
- HA adults had more frequent ER visits (p=0.02) than HB adults.
- Both adults (p<0.01) and children (p=0.04) with severe HA had higher annual factor usage than HB counterparts.
- There were no differences found among children with HA versus HB in ABR, hospitalizations, or ER visits.

CONCLUSIONS

- HUGS V HA and HB cohorts shared similar socio-demographic characteristics but displayed differing clinical outcomes, treatment practices and levels of healthcare utilization across disease severities and age groups.
- These findings add to the body of evidence that HB may be less clinically severe than HA.

ACKNOWLEDGEMENTS

We thank our sponsors: Baxter Healthcare Corporation, Bayer Foundation, CSL Behring, Novo Nordisk for funding the HUGS Va project and Pfizer (formerly Wyeth) for funding the HUGS Va and Vb projects.
The Hemophilia Utilization Group Studies Part V (HUGS V) (ranked by study center ID):
University of Southern California: Michael B. Nichol, PhD (Principal Investigator), Kathleen A. Johnson, PharmD MPH, PhD (late Principal Investigator), Mimi Lou, MS, Joanne Wu, MS, Zheng-Yi Zhou, PhD, Jiat Ling Poon, PhD, Xiaoli Niu, Christina Chen, PhD, Jason N. Doctor, PhD;
Children's Hospital Los Angeles, Hemostasis and Thrombosis Center: Cathliyn Buranahirun, PsyD (Site Principal Investigator), Robert Miller, PA (former Site Principal Investigator), Jennifer Hanley, Wendy Leung;
Children's Hospital of Orange County, Hemophilia Treatment Center: Amit Soni, MD (Site Principal Investigator), Heather Huszti, PhD (former Site Principal Investigator), James Fabella;
University of Colorado Denver: Brenda Riske, MS, MBA, MPA (Site Principal Investigator), Julie Smith, Kristi Norton;
Indiana Hemophilia & Thrombosis Center, Hemophilia Treatment Center: Amy Shapiro, MD (Site Principal Investigator), Natalie Duncan, MPH, Melissa Meyer, Brandy Trawinski, Jayme Harvey, Adrianna Williamson;
UMASS Memorial Hospital, New England Hemophilia Center: Ann D. Forsberg, MA, MPH (Site Principal Investigator), Patricia Forand;
University of Texas at Houston, Gulf States Hemophilia and Thrombophilia Center: Megan M. Ullman, MA, MPH (Site Principal Investigator);
Akron Children's Hospital Medical Center, Ohio: Jeffrey Hord, MD (Site Principal Investigator), Erin Cockrell, DO (former Site Principal Investigator), Dawn Ali, Felicia Lewis, Janice Kakish, Jill Bradisse;
Michigan State University, Center for Bleeding and Clotting Disorders: Roshni Kulkarni, MD (Site Principal Investigator), Laura Carlson, Sue Adkins;
BloodworksNW (formerly Puget Sound Blood Center) and the University of Washington: Barbara A. Konkle, MD (Site Principal Investigator), Sarah Ruuska, Sarah Galdzicka;
University of Mississippi Medical Center: Suvankar Majumdar, MD (Site Principal Investigator), Linnea McMillan.
The authors thank the **HUGS Steering Committee:** Randall G. Curtis, MBA, Shelby L. Dietrich, MD and Marion A. Koerper, MD, Brenda Riske, MS, MBA, MPA, Megan M. Ullman, MA, MPH, Judith R. Baker, DrPH, MHSA, and **Consultants** Femida Gwadry-Sridhar, BSPhm, MS, PhD, Jason N. Doctor, PhD and Kathy Parish, PhD.
Corresponding author: Michael B. Nichol, mnichol@usc.edu. Dr. Nichol is a consultant for Biogen and Bayer.

PosterSessionOnline.com
Poster
Presented at:
WFH 2016 WORLD CONGRESS
DOI: 10.3252/pso.eu.WFH2016.2016
Outcome Assessment
Mimi Lou
121--PP-W
WFH2016