Poster 106-PP-W

The Application of Thrombin Generation Assays Is Compromised by Low Levels of Antithrombin III

Knappe S., Till S., Ullrich N., Gerstenbauer G., Scheiflinger F., Dockal M.

Shire, Vienna, Austria

INTRODUCTION

- Antithrombin III (ATIII) is a stoichemetric inhibitor of thrombin and factor Xa and is crucial for the regulation of coagulation.
- A decrease in ATIII causes a rise in active thrombin. For example, a reduction of ATIII in patients with congenital ATIII deficiency causes a rise of active thrombin and the patients are at risk for venous and atrial thrombosis (1,2).
- ATIII reduction has been considered as a treatment for hemophilia A, a congenital bleeding disorder.
- To monitor thrombin generation in human plasma, the calibrated automated thrombogram (CAT) assay is widely used.
- The test is based on fluorescence that is generated by cleavage of the thrombin-specific substrate Z-G-G-R-AMC.
- Fluorescence signals from sample and calibrator (alpha-2-macroglobulin thrombin complex) are processed to calculate the amount of thrombin, provided the substrate is in excess.

OBJECTIVE

- We tested the technical feasibility of assessing the effect of ATIII reduction on thrombin generation in human plasma.

METHODS

- Thrombin generation was evaluated by CAT, a method described by Hemker et al. (3). For quantification of thrombin at each time point, a thrombin calibrator was included for each plasma sample.
- To study the effect of ATIII on thrombin generation, ATIII-deficient plasma (Affinity Biologicals) was used. First, plasma was treated with corn trypsin inhibitor (final conc. 41.3 µg/mL; Haematologic Technologies) to inhibit undesired contact activation by factor XIIa. Optionally, factor VIII (FVIII) was inhibited by adding 50 BU/mL goat anti-FVIII (Baxalta). Increasing concentrations of purified plasma ATIII (0-2.5 µM; Enzyme Research Laboratories) were spiked into plasma.
- Thrombin generation was triggered with 1 pM tissue factor (TF) and 4 µM phospholipids (PPP LOW; Thrombinoscope). Samples were recalcified by FluCa reagent containing the substrate and $CaCl_2$ (Thrombinoscope). Fluorescence was measured in a Fluoroskan Ascent® plate reader (ThermoScientific; filters 390 nm excitation and 460 nm emission) at 37°C for 90 min. All measurements were performed in duplicate and the assay repeated 3 times.
- Thrombin levels were calculated using the calibrator, and parameters of resulting thrombin generation curves were evaluated via Thrombinoscope software.(Figure 1).

RESULTS

Figure 1: Calibrated automated thrombogram- Principle and parameters

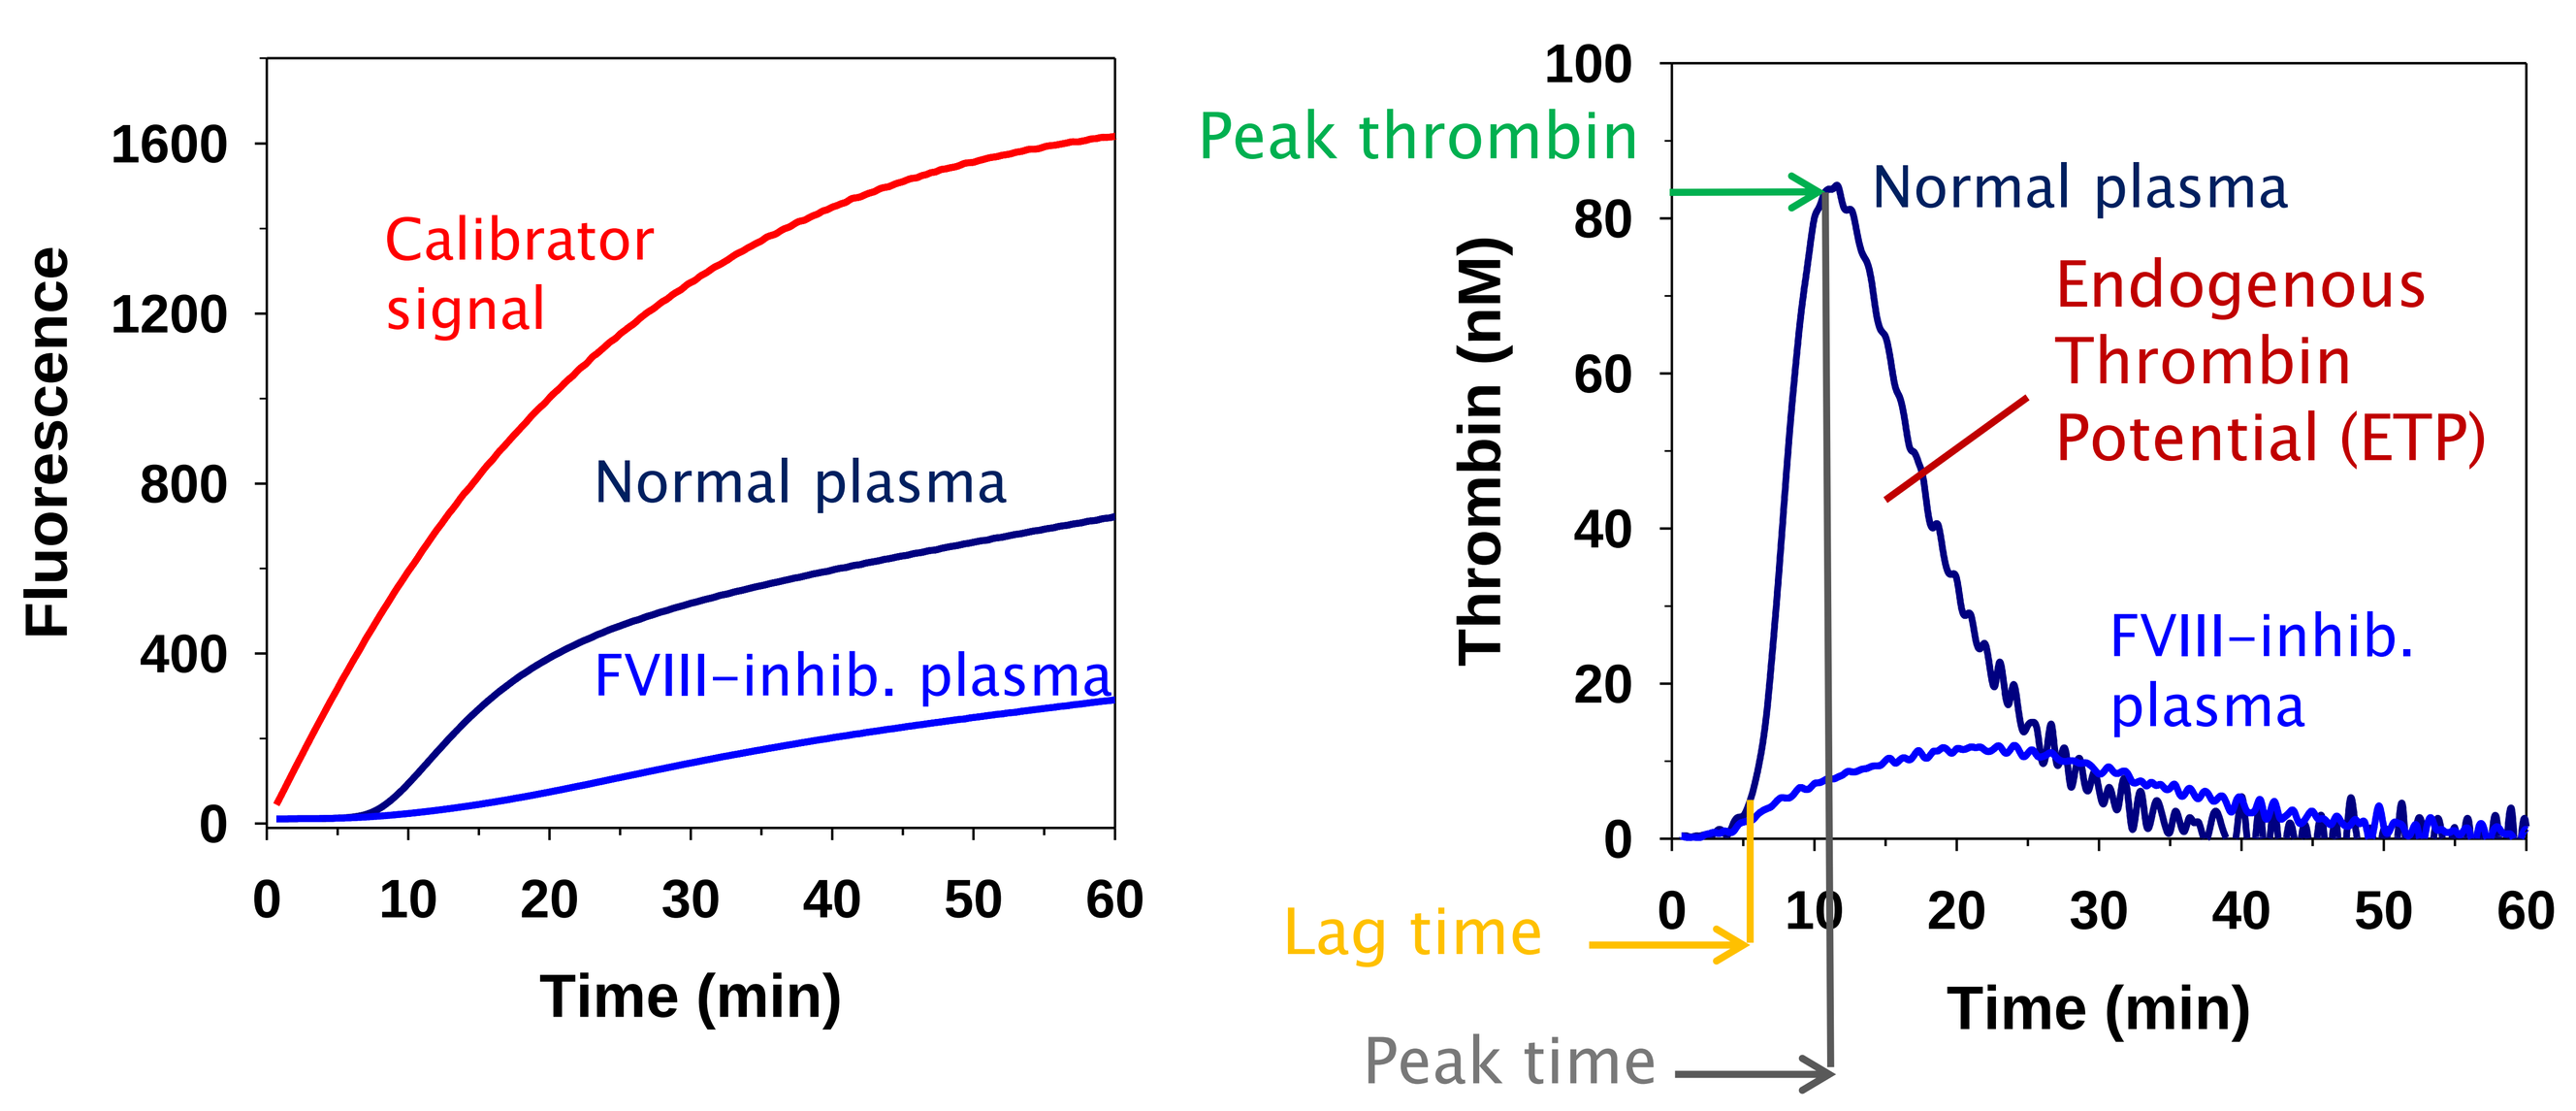
Fluorescence
1600
1200
800
400
0
Calibrator signal
Normal plasma
FVIII-inhib. plasma
0 10 20 30 40 50 60
Time (min)
Thrombin (nM)
100
80
60
40
20
0
Peak thrombin
Normal plasma
Endogenous Thrombin Potential (ETP)
FVIII-inhib. plasma
Lag time
Peak time
Time (min)

Representative fluoresence signals of calibrator, a normal and a FVIII-inhibited (hemophilia A conditon) human plasma sample. (left panel). CAT parameters : lag time (the initiation of thrombin generation), the peak time at which the maximum thrombin amount (peak thrombin) is reached, and the endogenous thrombin potential (ETP) that represents the area under the curve (right panel).

Figure 2: Fluorescence signal for plasma with and without ATIII

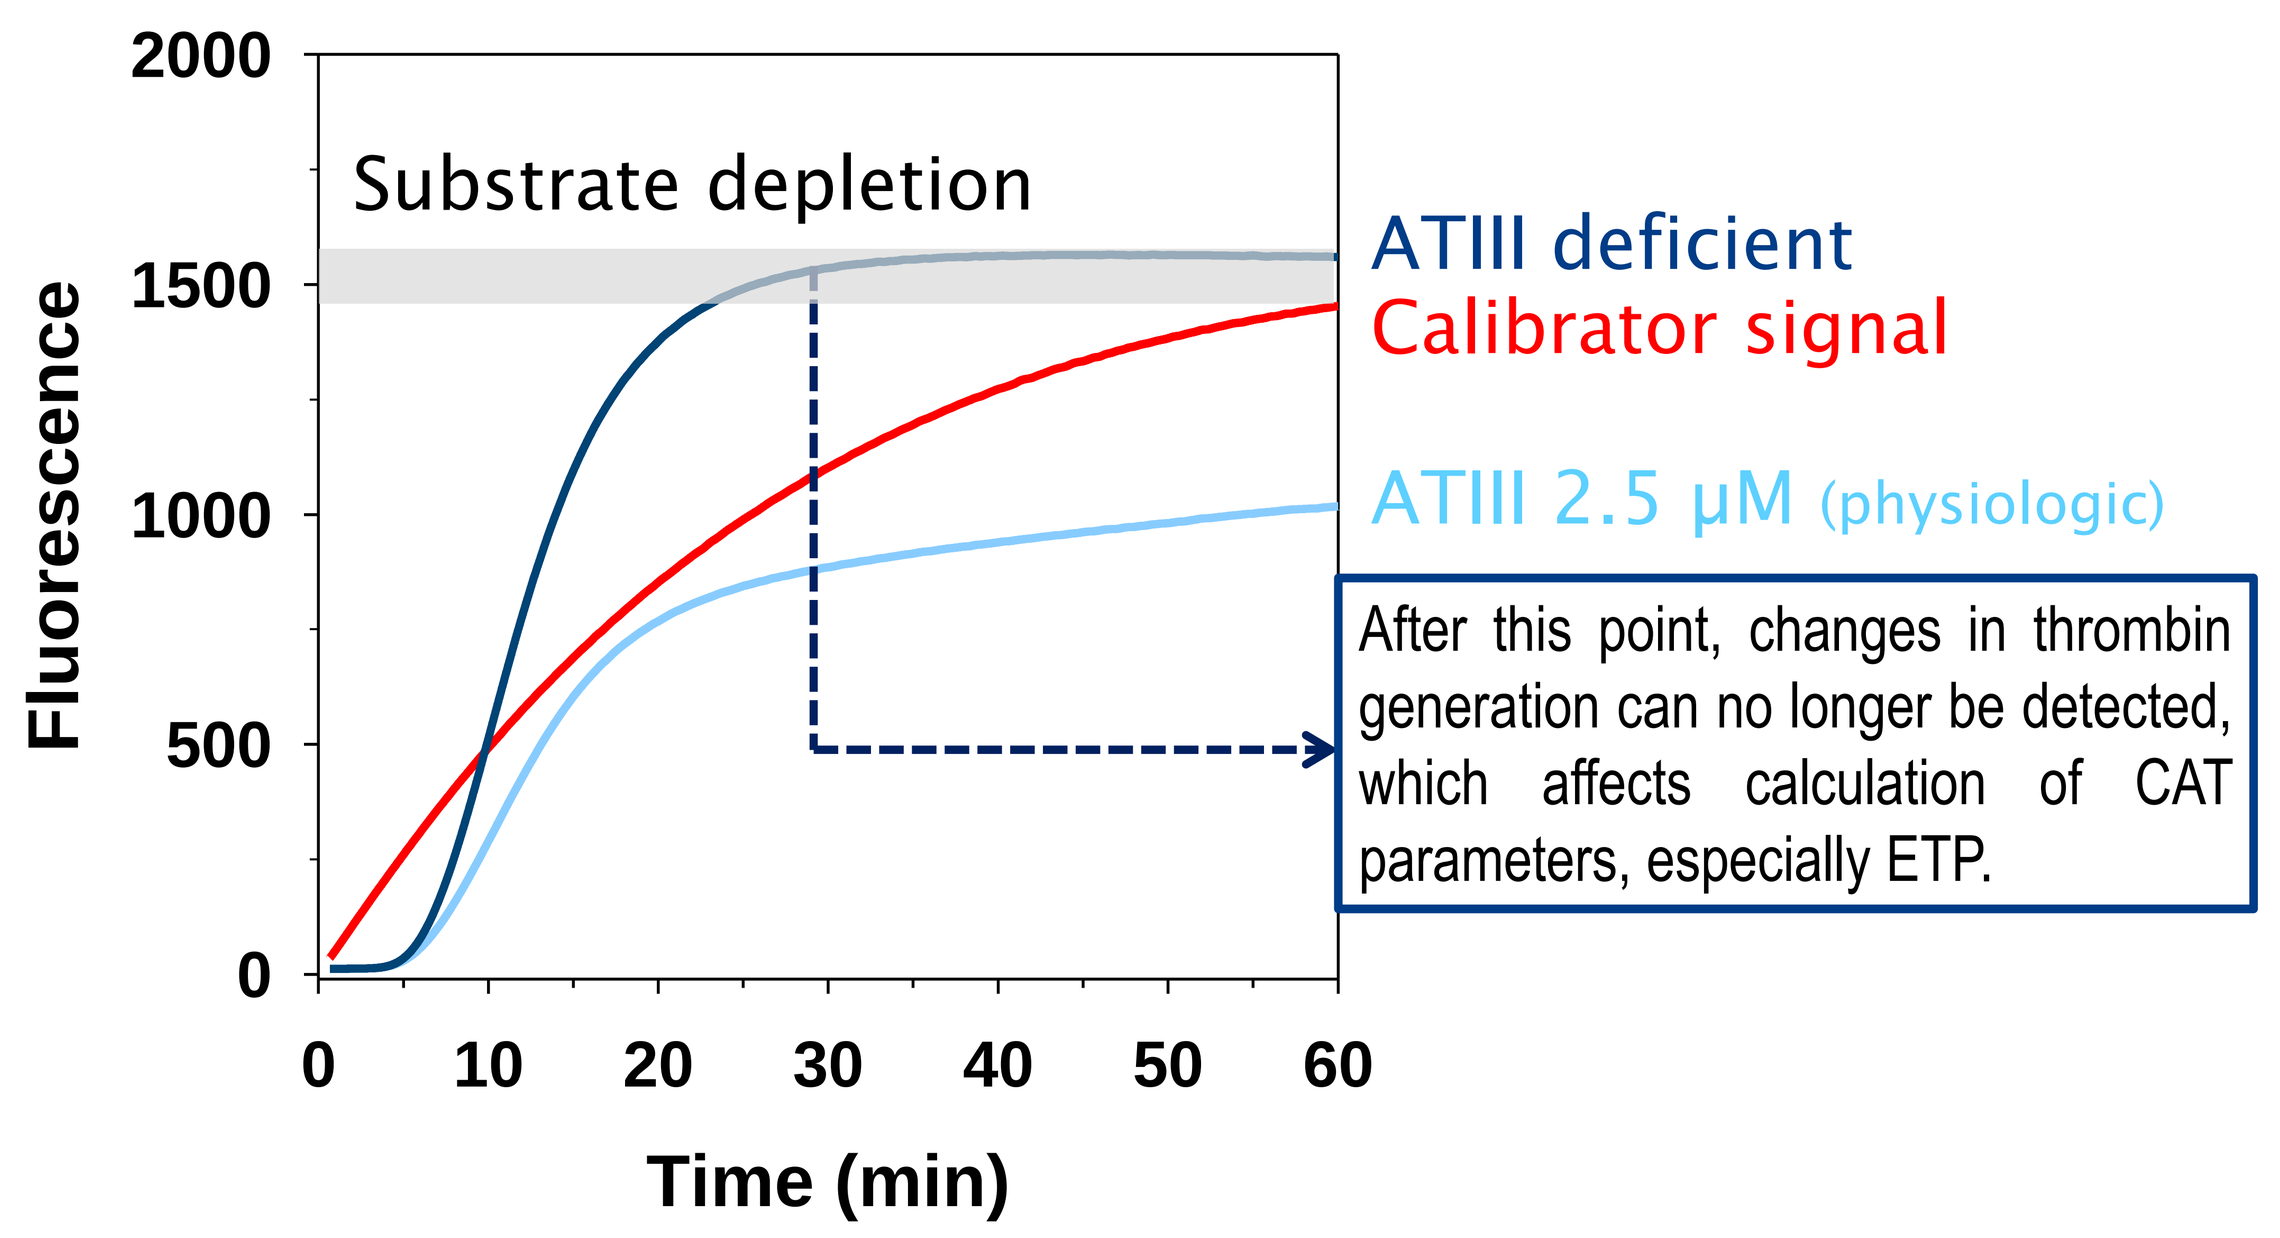
2000
1500
1000
500
0
Fluorescence
Substrate depletion
ATIII deficient
Calibrator signal
ATIII 2.5 µM (physiologic)
After this point, changes in thrombin generation can no longer be detected, which affects calculation of CAT parameters, especially ETP.
0 10 20 30 40 50 60
Time (min)

- Tissue factor triggered CAT was performed in ATIII-deficient plasma with or without supplementation with 2.5 µM ATIII, the physiological ATIII plasma concentration.
- In the absence of ATIII, the fluorescent substrate is consumed rapidly, leading to substrate depletion .
- In samples without ATIII, raw data analysis showed that the fluorescence signal reached a plateau after ~30 min.

Figure 3: Thrombin generation at reduced ATIII levels in presence of FVIII

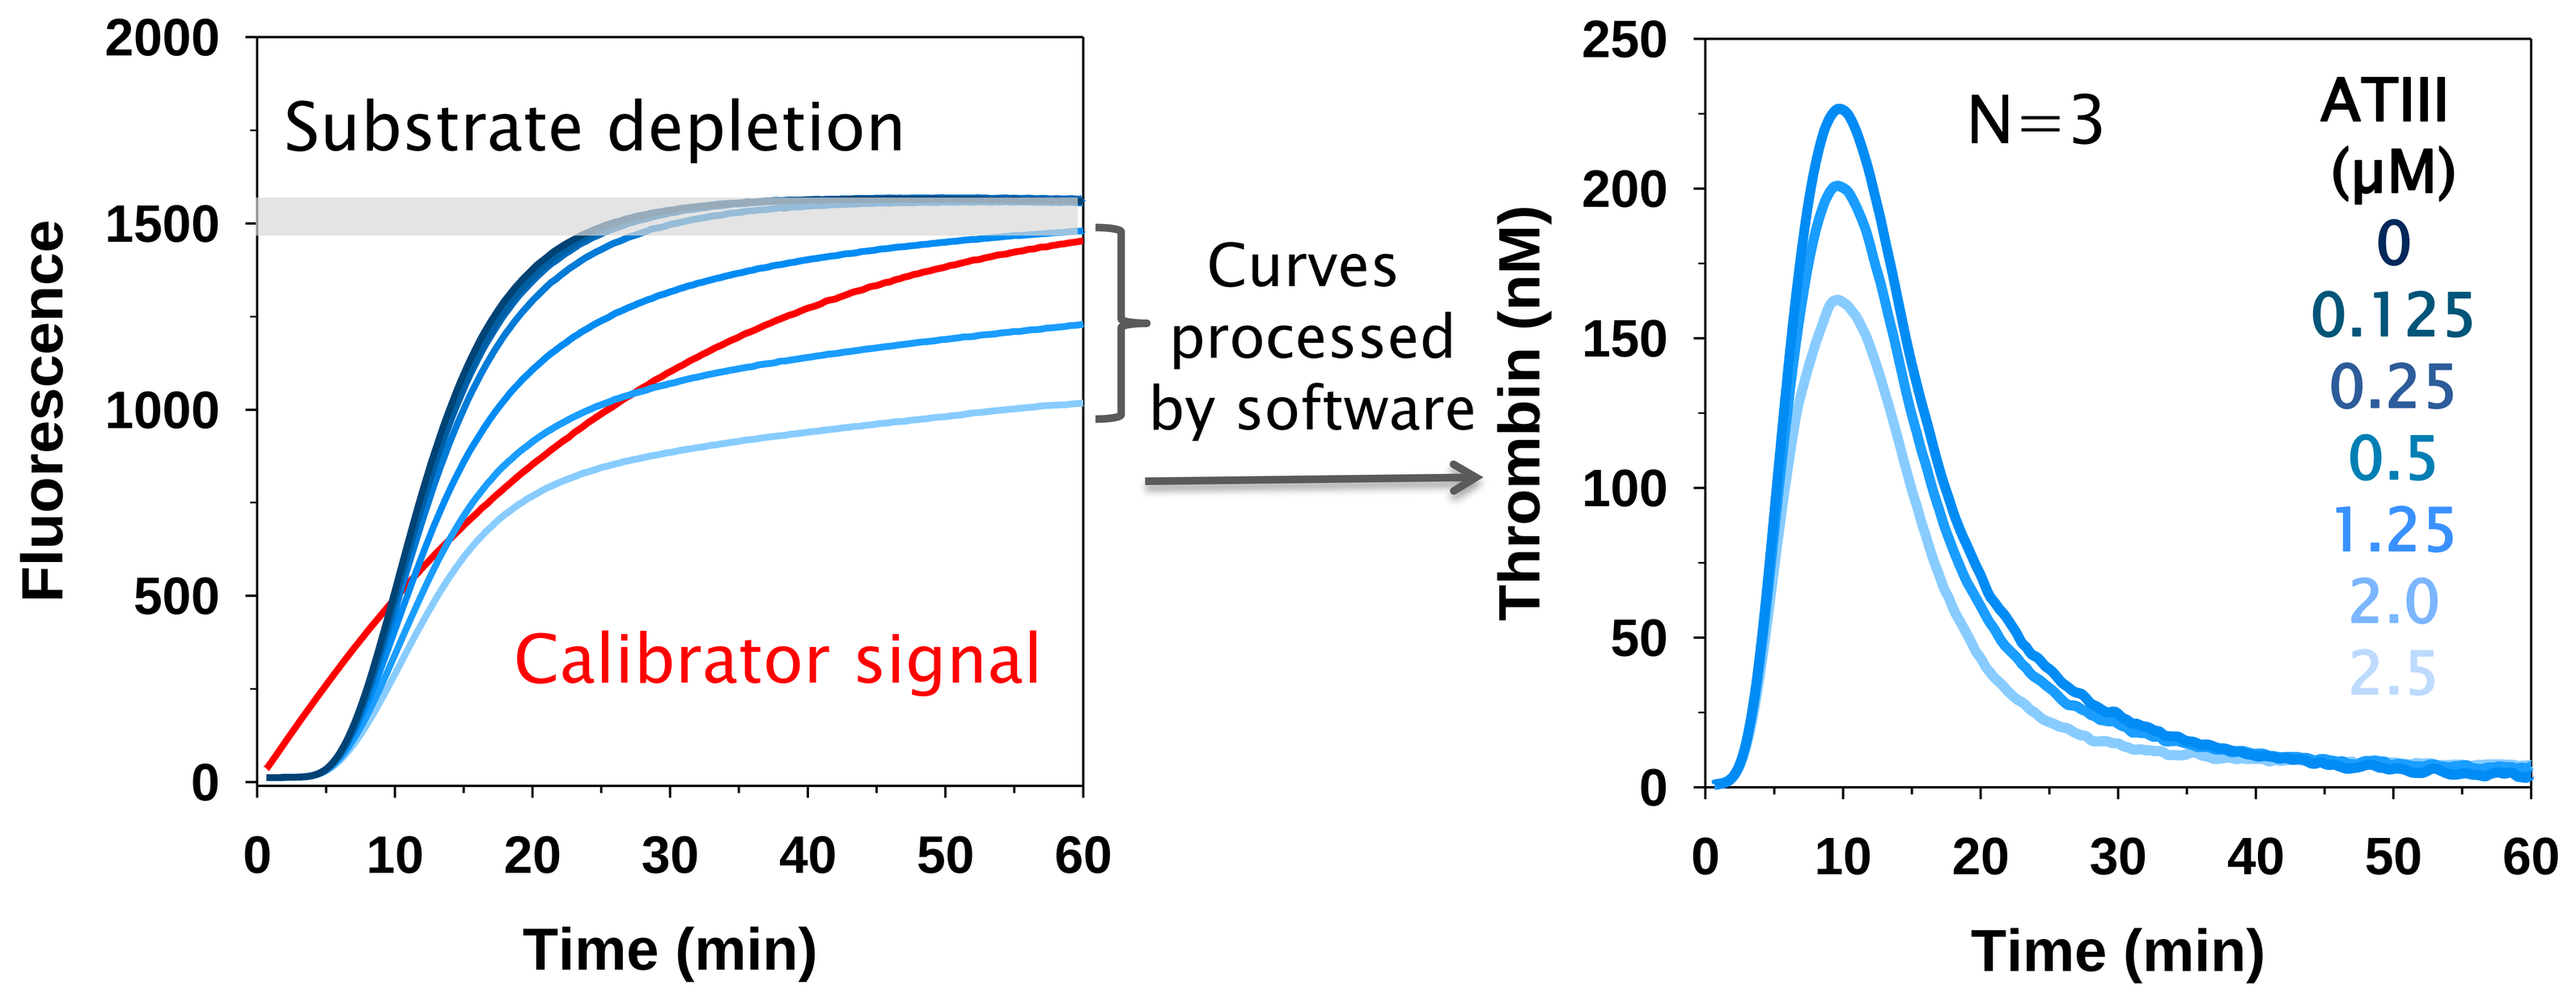
Fluorescence
2000
1500
1000
500
0
Substrate depletion
Calibrator signal
0 10 20 30 40 50 60
Time (min)
Curves processed by software
Thrombin (nM)
250
200
150
100
50
0
N=3
ATIII (µM)
0
0.125
0.25
0.5
1.25
2.0
2.5
Time (min)

Tissue factor triggered CAT in ATIII-deficient plasma in the presence of factor VIII: Fluorescence signals of calibrator and plasma samples without ATIII or supplemented with 0.125 – 2.5 µM ATIII (left panel). Respective thrombin generation profiles for plasma samples with 1.25 - 2.5 µM ATIII as processed by the instrument's software. The samples with 0 – 0.5 µM ATIII could not be processed by the software (right panel).

- Tissue factor triggered CAT was performed in ATIII-deficient plasma in the presence of factor VIII.
- Plasma with ≥ 1.25 µM ATIII (50%) is not affected by substrate depletion. Only under these conditions is the software able to process all CAT parameters.

Figure 4: Thrombin generation at reduced ATIII levels in absence of FVIII (hemophilia A condition)

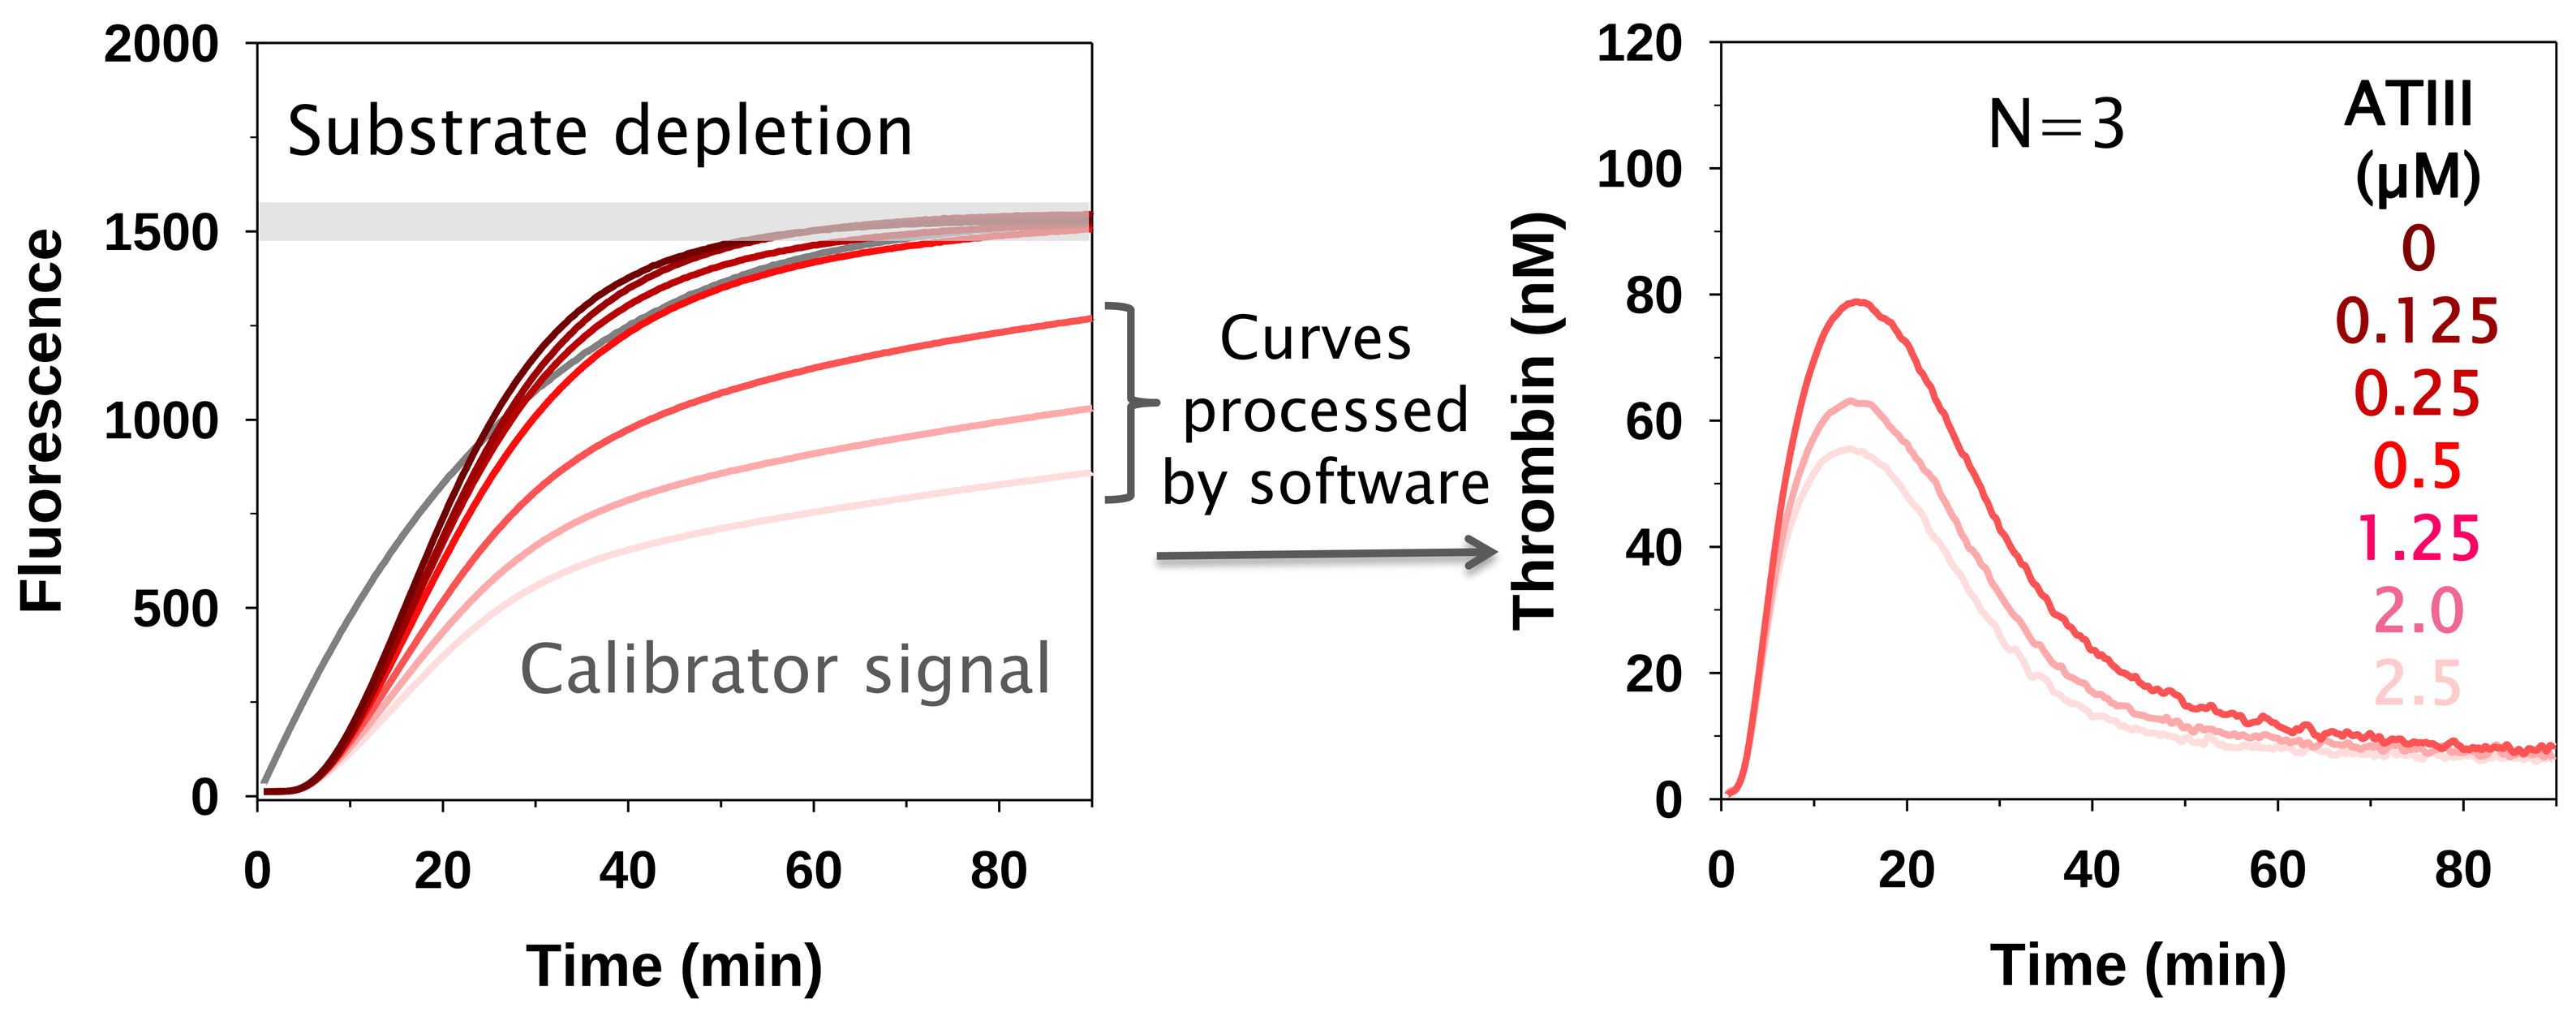
Fluorescence
2000
1500
1000
500
0
Substrate depletion
Calibrator signal
0 20 40 60 80
Time (min)
Curves processed by software
Thrombin (nM)
120
100
80
60
40
20
0
N=3
ATIII (µM)
0
0.125
0.25
0.5
1.25
2.0
2.5
Time (min)

Tissue factor triggered CAT in ATIII-deficient plasma in the absence of factor VIII: The assay was performed in the presence of anti-human FVIII antibodies to simulate hemophilia A conditions. Fluorescence signals of calibrator and plasma samples without ATIII or supplemented with 0.125 – 2.5 µM ATIII (left panel). Respective thrombin generation profiles for plasma samples with 1.25 - 2.5 µM ATIII as processed by the instrument's software. The samples with 0 – 0.5 µM ATIII could not be processed by the software (right panel).

- Tissue factor triggered CAT was performed in ATIII-deficient plasma in the presence of an anti-human FVIII antibody used to simulate hemophilia A conditions.
- Even under conditions where coagulation is reduced due to the FVIII inhibition, substrate depletion occurs after ~50 min.

CONCLUSION

- Thrombin generation at low ATIII plasma levels by CAT leads to rapid consumption of fluorogenic substrate.
- It is impossible to distinguish between thrombin regulation by ATIII and excessive substrate consumption.
- This limitation occurs at ATIII plasma levels of ≤ 50% of normal plasma, and can lead to underestimation of ETP values.
- *Ex vivo* thrombin generation resulting from therapeutic interventions targeting ATIII is most certainly compromised at substantially reduced ATIII plasma concentrations.

REFERENCES

1. Duchemin et al., 2008 Thromb Haemost; 99:767-773
2. Dielis et al., 2008 J Thromb Haemost; 6:125-131
3. Hemker et al., 2003 Pathophys Haemos Thromb; 33: 4-15

DISCLOSURES

All authors are full-time employees of Shire, Vienna, Austria

WFH World Congress; July 24–28, 2016; Orlando, USA

PosterSessionOnline.com
Poster Presented at:
DOI: 10.3252/pso.eu.WFH2016.2016
Laboratory issues
Michael Dockal
106--PP-W
WFH2016