P-W-105

Results from a multi-national survey of FVIII activity assay preferences

Peter L. Turecek, Claudia Apostol, Herbert Gritsch

Shire, Vienna, Austria

INTRODUCTION

Historically clinical laboratories used one-stage clotting assays for FVIII activity determination for diagnosis of hemophilia, determination of the phenotypic severity of FVIII deficiency, and to monitor post-infusion FVIII levels to adjust dosing and nowadays also to personalize treatment regimes. Based on literature and feedback from experts it is assumed that the vast majority of laboratories still use one-stage clotting assays while chromogenic assays are rarely used, and if, mainly in addition to one-stage clotting assays to confirm results and/or refine analysis, particularly for diagnosis.

OBJECTIVE

A multi-national survey of FVIII activity assay practice was performed to investigate current FVIII activity assay preferences. Here we report the results of the survey.

METHODS

A questionnaire was sent to 127 laboratories in 25 countries in all geographies. The questionnaire asked for details on assay methods, instruments, assay reagents including FVIII deficient plasmas and aPTT reagents, reference standards, frequency of preparation of reference curve, number of different dilutions tested per sample, assay controls, and further details on patient plasma samples testing. Feedback was collected and the answers were evaluated.

RESULTS

56 laboratories returned the completed questionnaire. From these laboratories, 98% perform and rely on the one-stage clotting assay, and 73% are using the one-stage clotting assay as their sole FVIII activity test system. Only one single laboratory (2%) uses the chromogenic assay alone, while 25% have both assays available

Figure 1: One-stage clotting and chromogenic assays in clinical laboratories

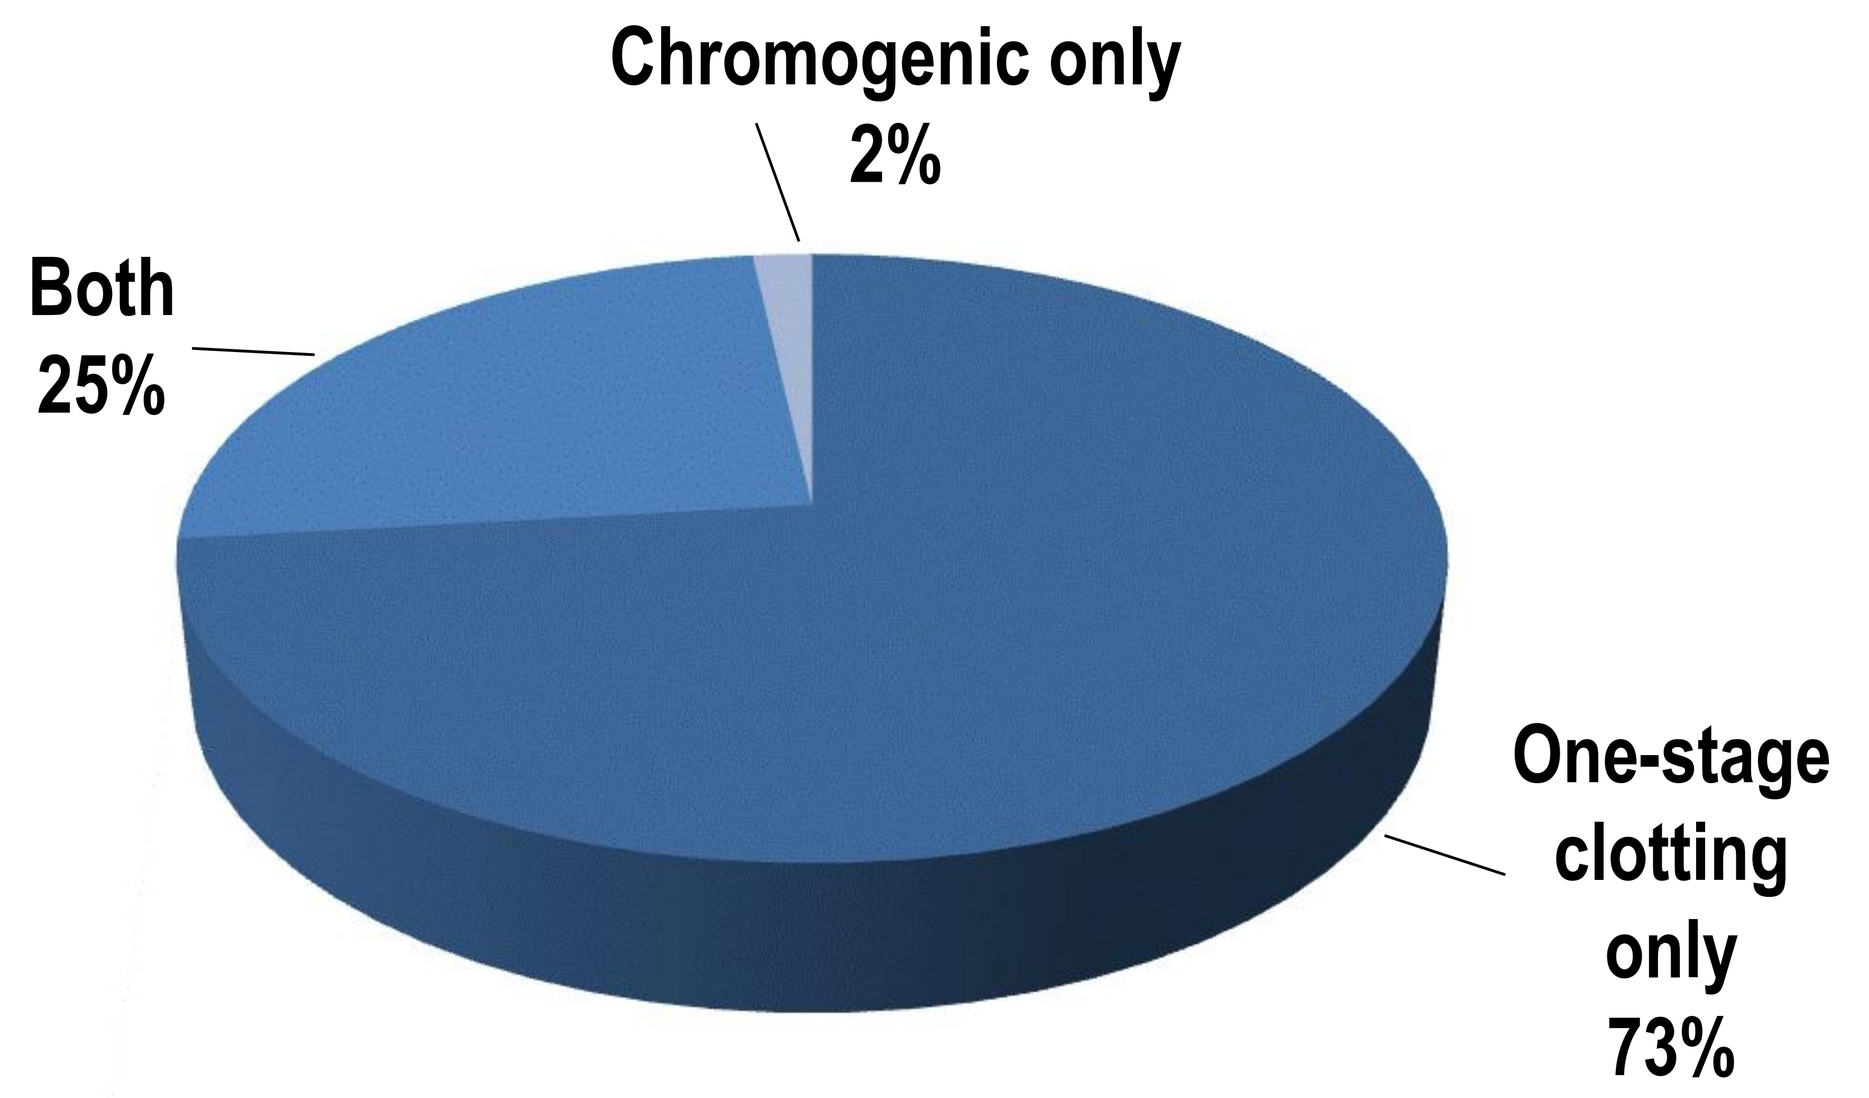
Chromogenic only 2%
Both 25%
One-stage clotting only 73%

REFERENCE

P.L. Turecek, S. Romeder-Finger, C. Apostol, A. Bauer, A. Crocker-Buque, D.A. Burger, R. Schall, H. Gritsch. A world-wide survey and field study in clinical hemostasis laboratories to evaluate FVIII:C activity assay variability of ADYNOVATE and OBIZUR in comparison to ADVATE. Haemophilia. 2016 Jun 28. doi: 10.1111/hae.13001. [Epub ahead of print].

WFH World Congress; July 24–28, 2016; Orlando, USA

RESULTS – ONE-STAGE CLOTTING ASSAYS

- From the laboratories performing the one-stage clotting assay as their preferred assay system, the majority (58%) use silica based aPTT reagents followed by ellagic acid based reagents. Kaolin activators and polyphenols are less frequently used. Coagulation analyzers from a variety of manufacturers are used, mainly instruments from Instrumentation Laboratories, Stago, Siemens and Sysmex.

Figure 2: Analytical instruments

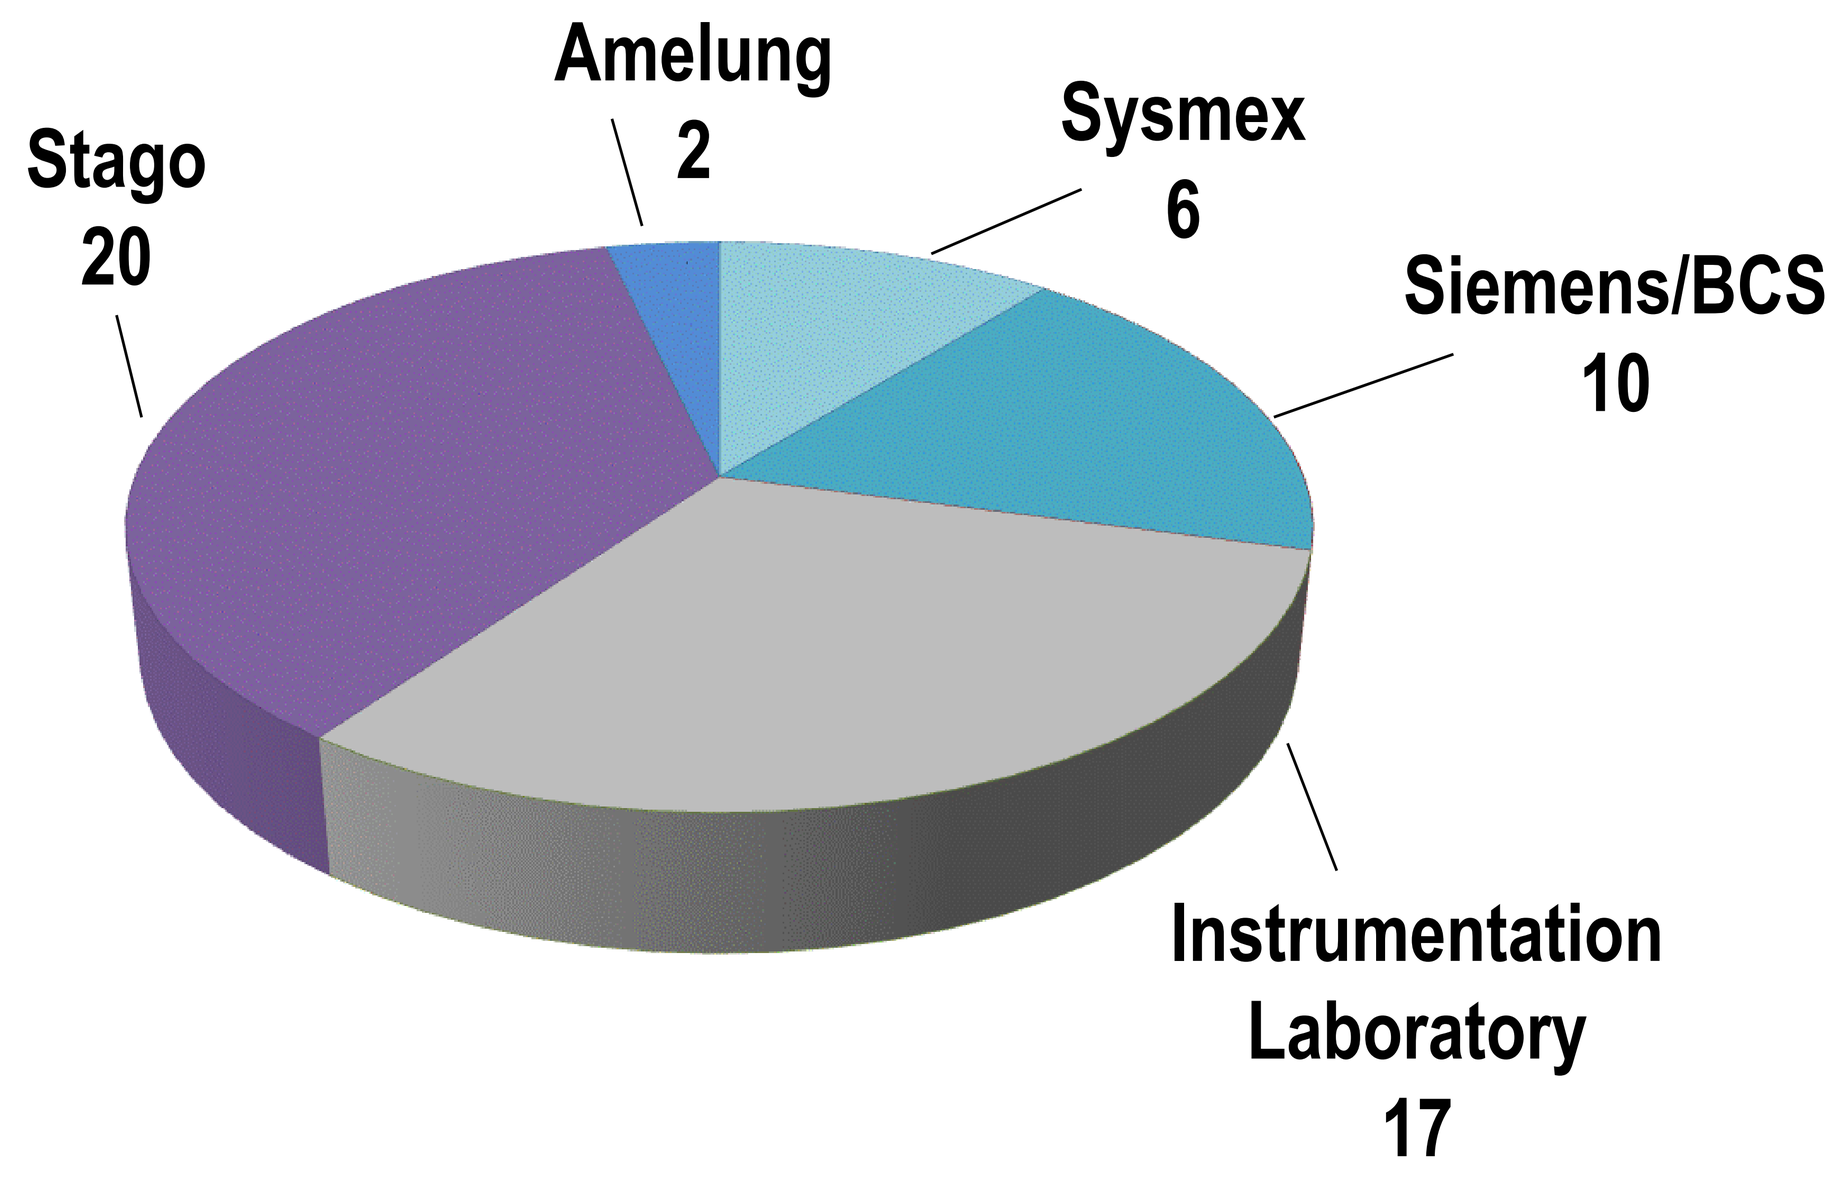
Amelung 2
Sysmex 6
Siemens/BCS 10
Instrumentation Laboratory 17
Stago 20

Figure 3: aPTT reagents for one-stage clotting assays

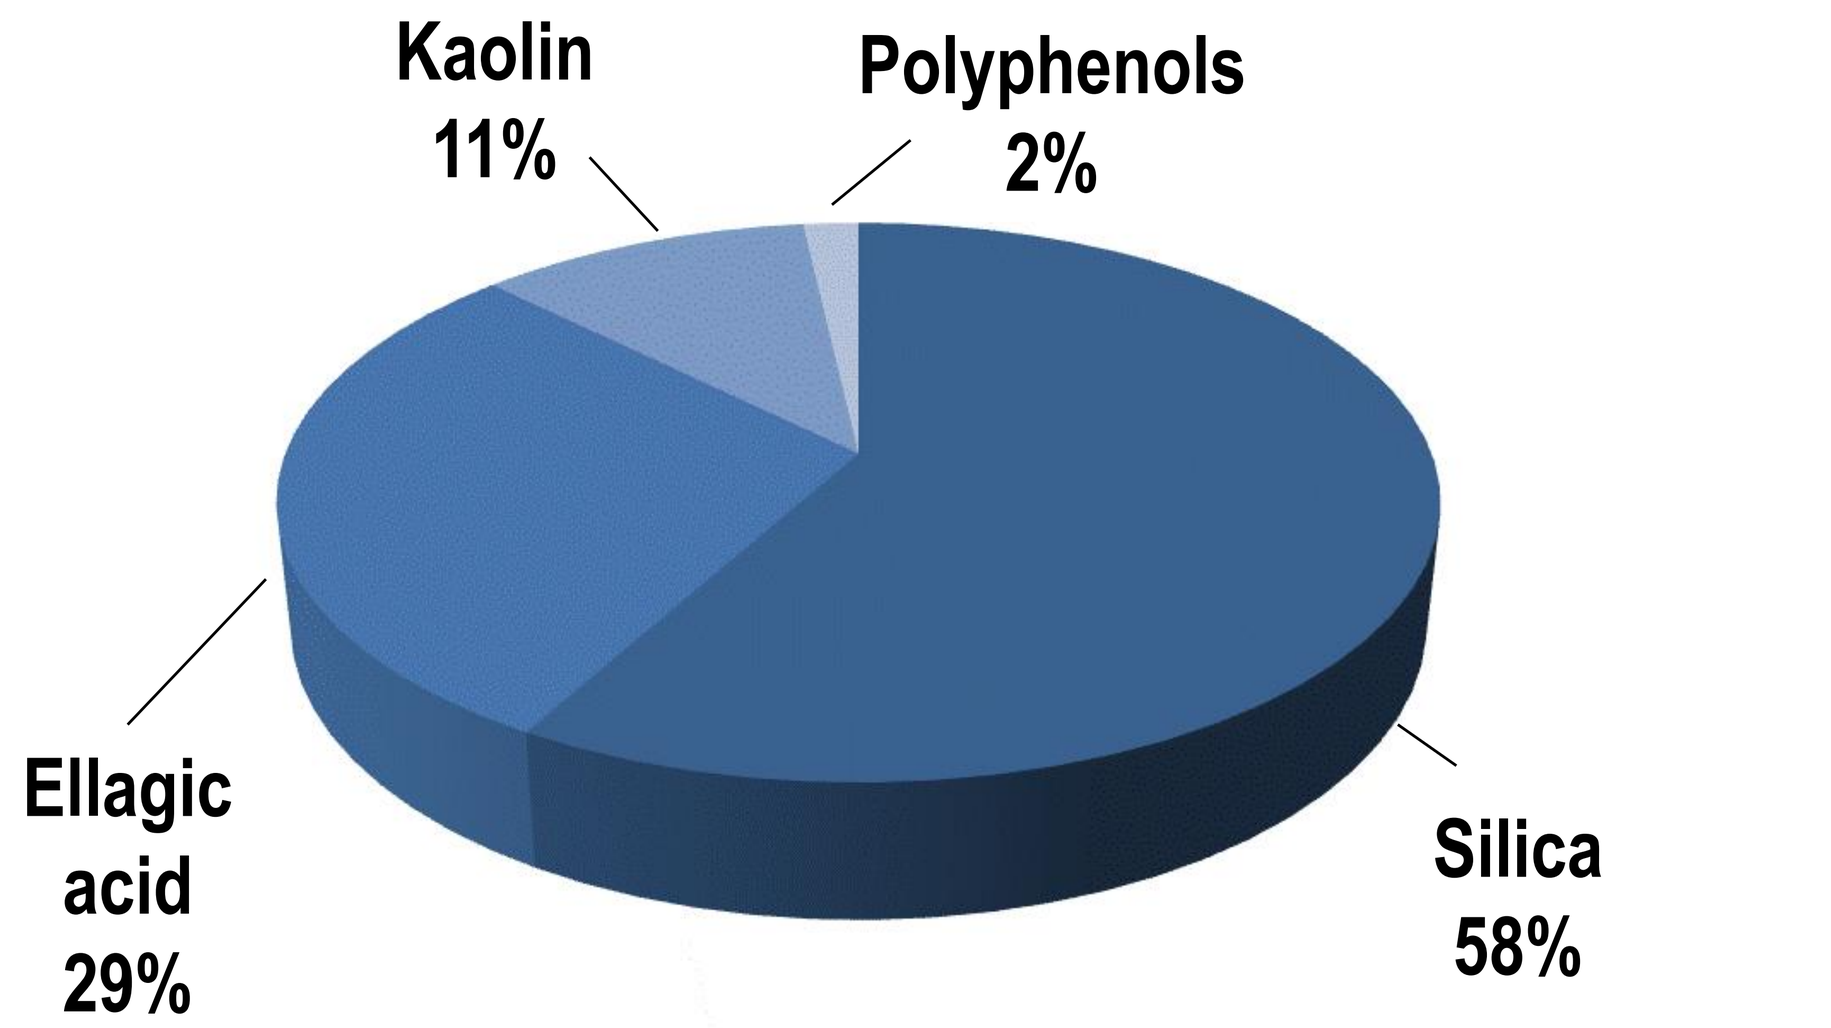
Kaolin 11%
Polyphenols 2%
Silica 58%
Ellagic acid 29%

- Reference standards for one-stage clotting assays are mainly from commercial source (Instrumentation Laboratories, Stago, Siemens, PrecisionBiologic, Roche), only one laboratory reported to use an in-house normal plasma pool, one provided no answer. The majority of laboratories relies on the reference calibration by the manufacturer (65%) while 33% perform an in-house calibration. Eighty percent (80%) of laboratories use immunodepleted FVIII-deficient plasma, 15% reported to use congenital hemophilia A plasma and 5% provided no answer.

Figure 4: Source of reference plasma

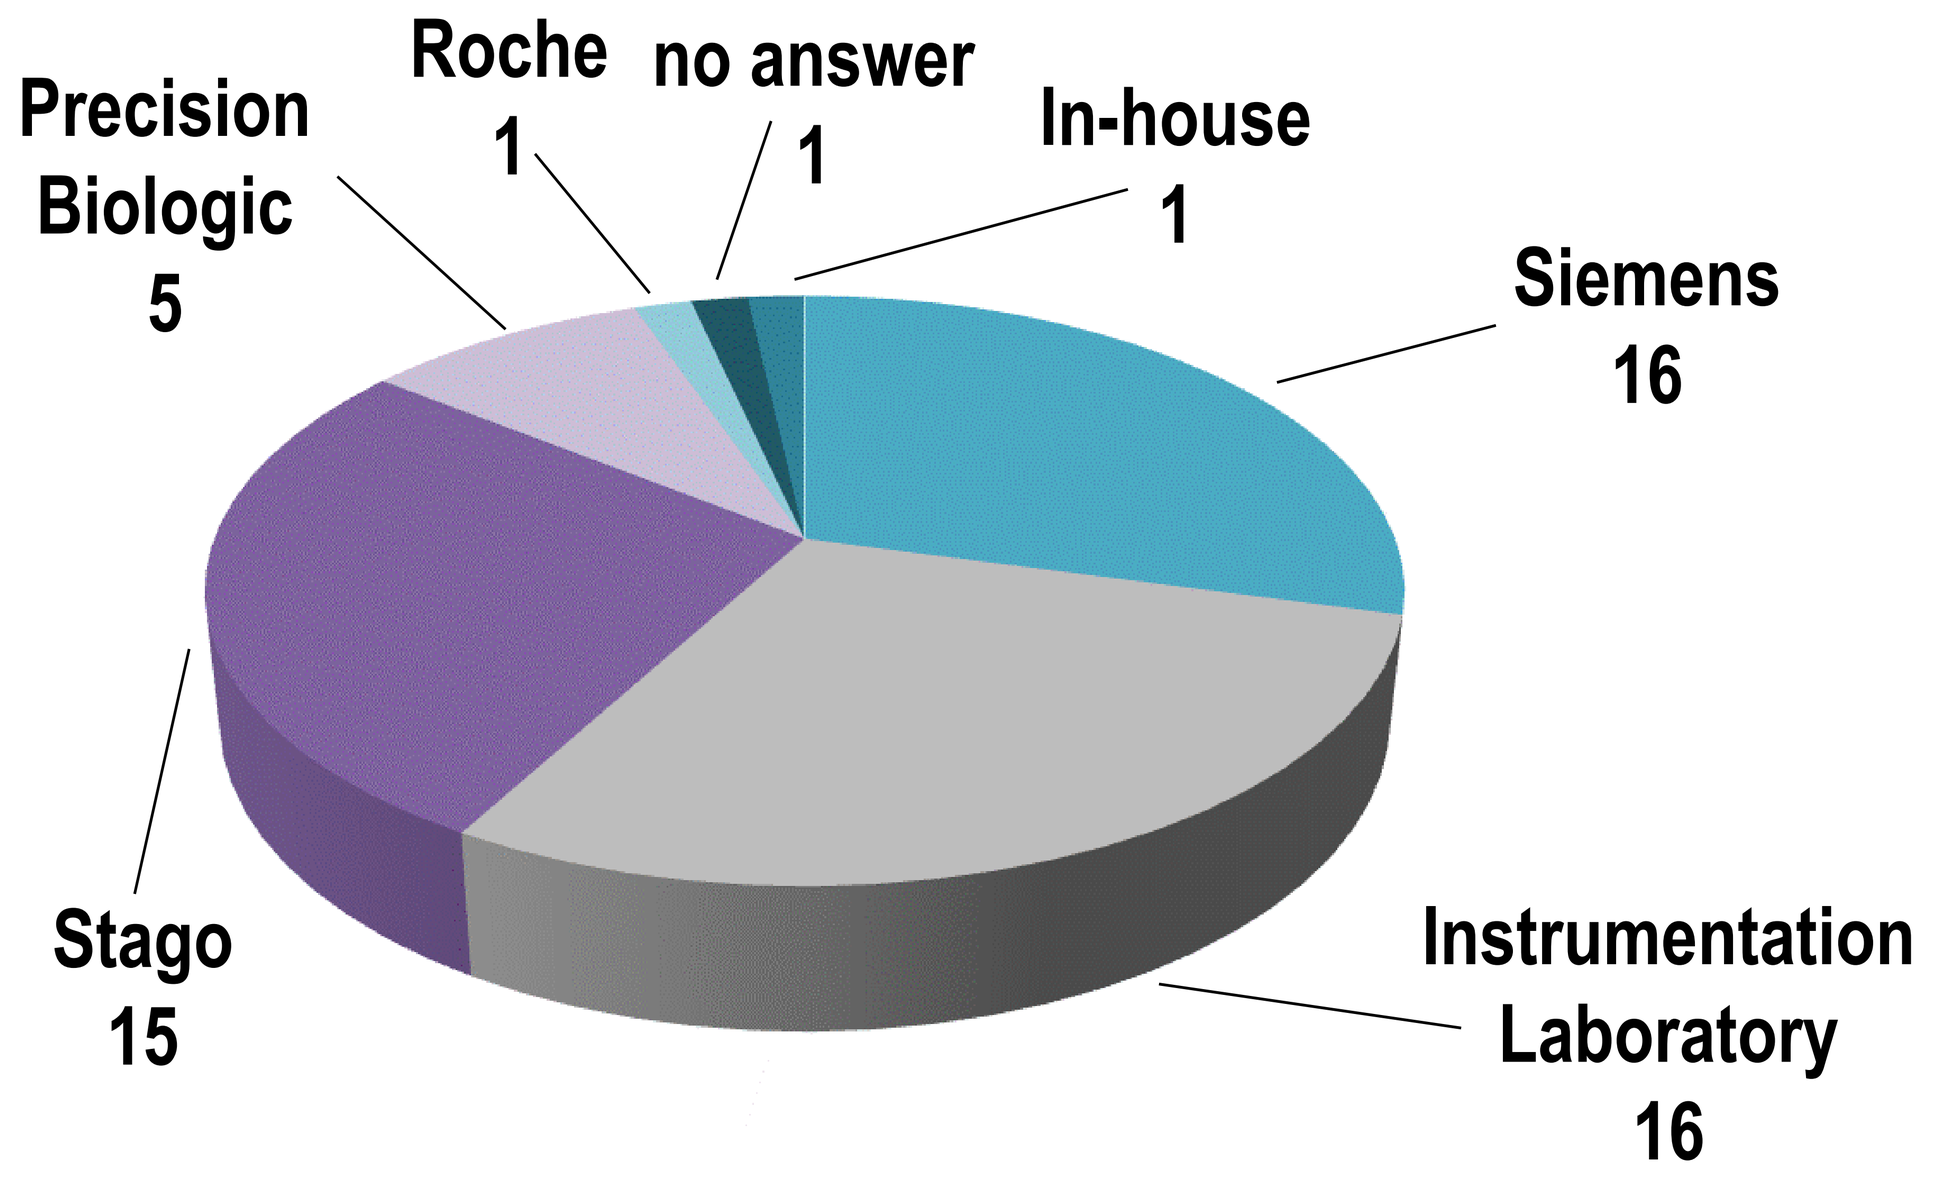
Precision Biologic 5
Roche 1
no answer 1
In-house 1
Siemens 16
Instrumentation Laboratory 16
Stago 15

Figure 5: FVIII-deficient plasma

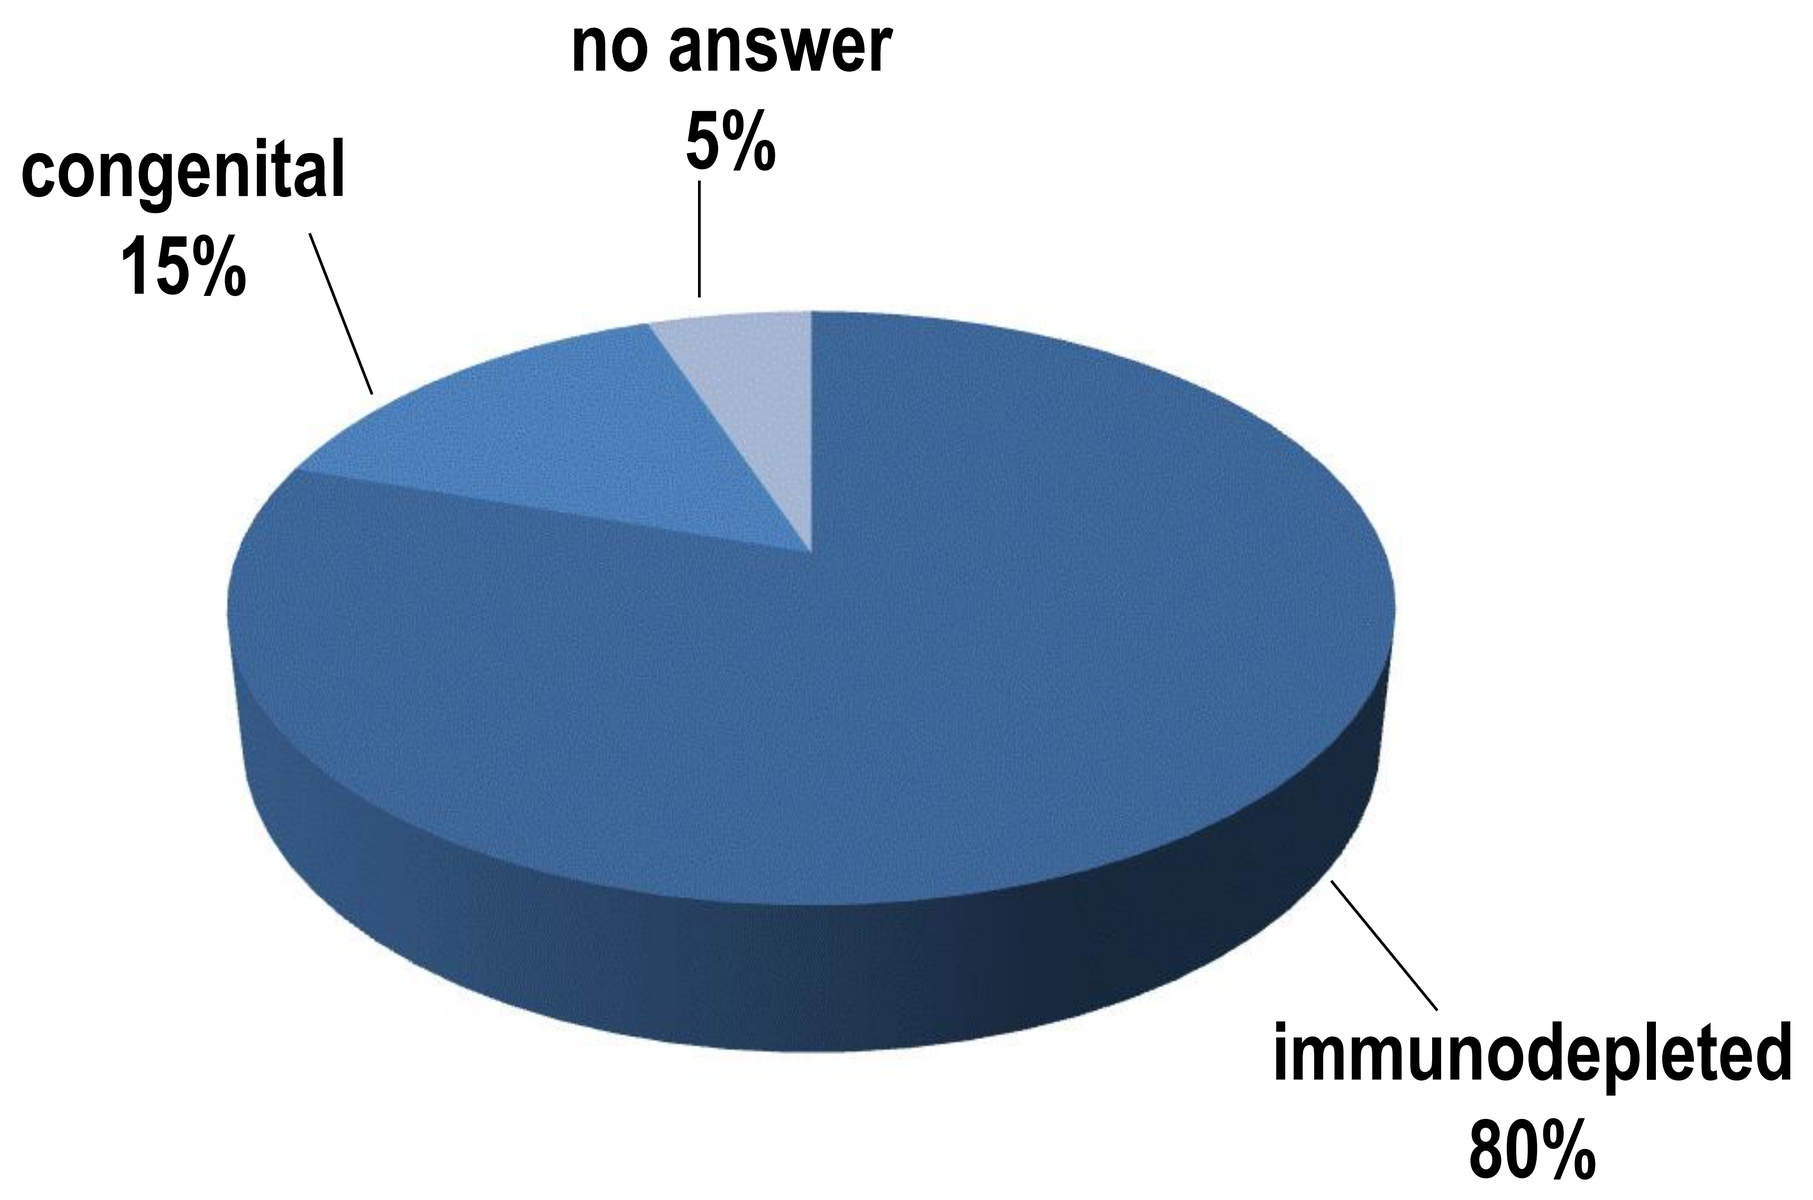
no answer 5%
congenital 15%
immunodepleted 80%

- Most of the laboratories use stored reference curves, replaced with every change in critical reagents (25). Some laboratories are running reference curves on a daily basis (12) or at other regular intervals (16). Patient samples are analyzed in 1 to 8 different dilutions, the majority performs 3 dilutions.

Figure 6: Frequency of reference curve generation

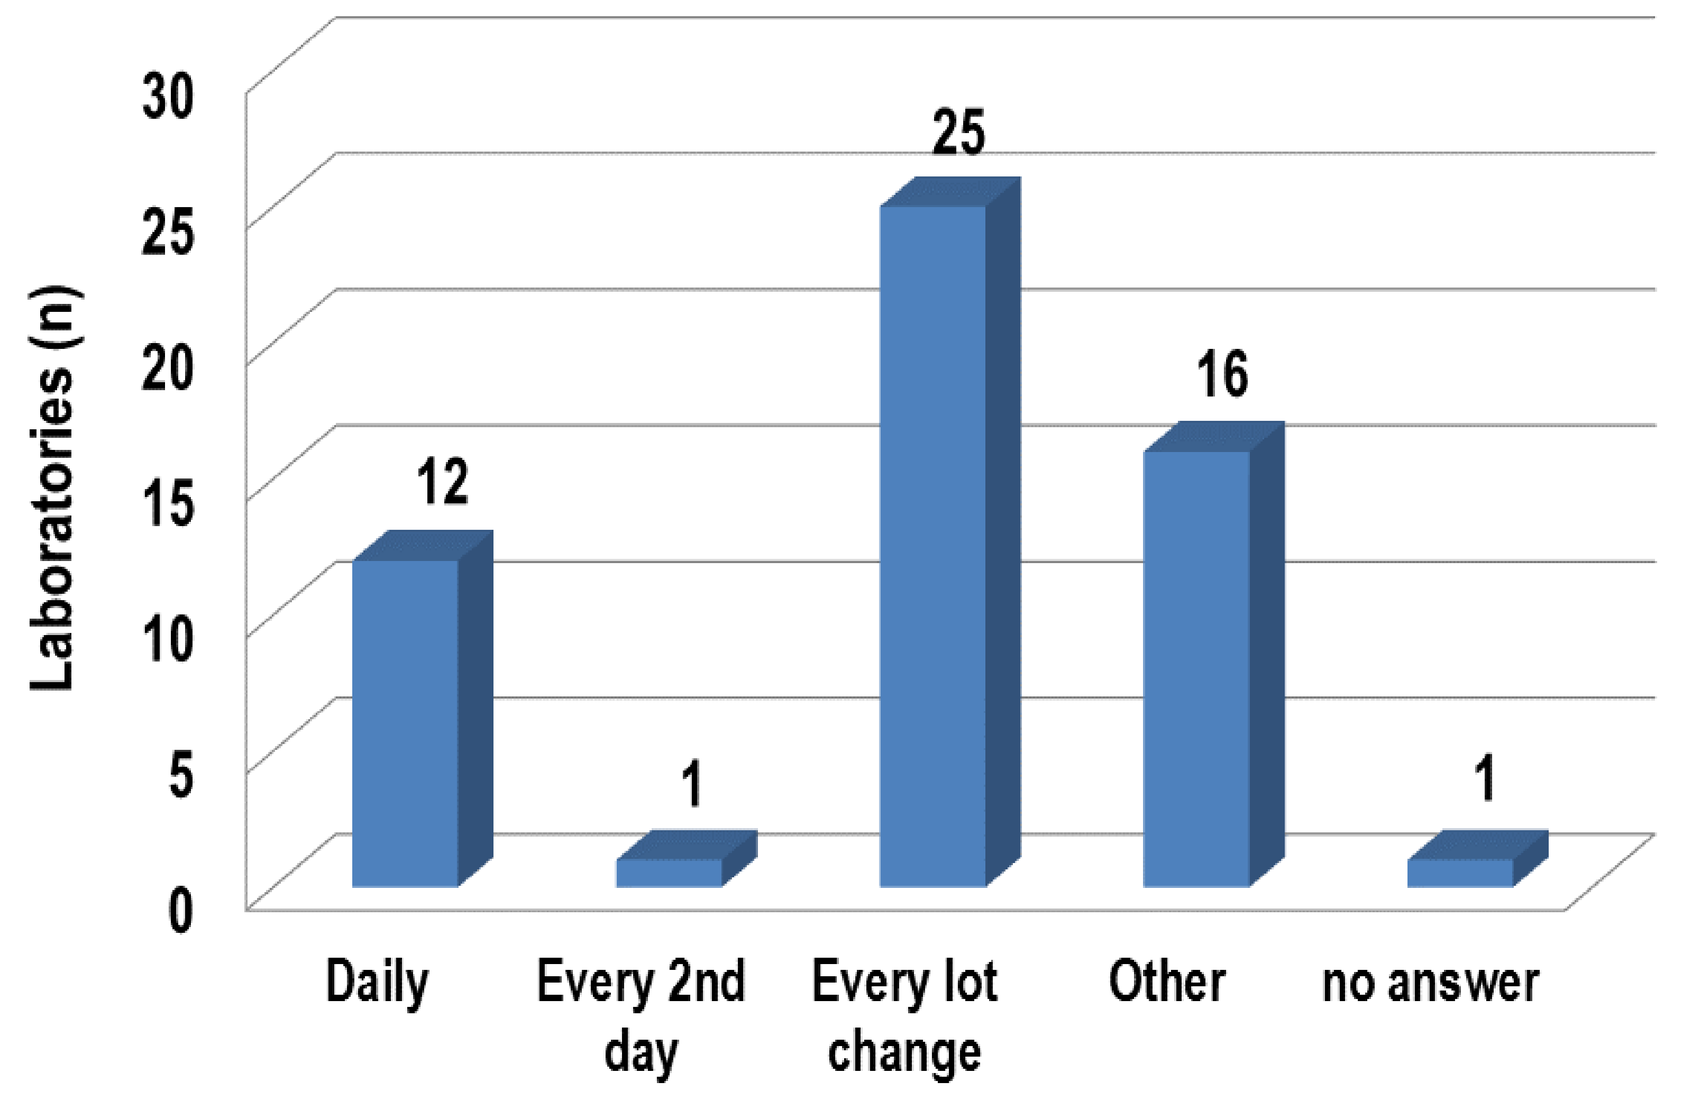
Laboratories (n)
0
5
10
15
20
25
30
12
1
25
16
1
Daily
Every 2nd day
Every lot change
Other
no answer

Figure 7: Patient samples, number of dilutions analyzed

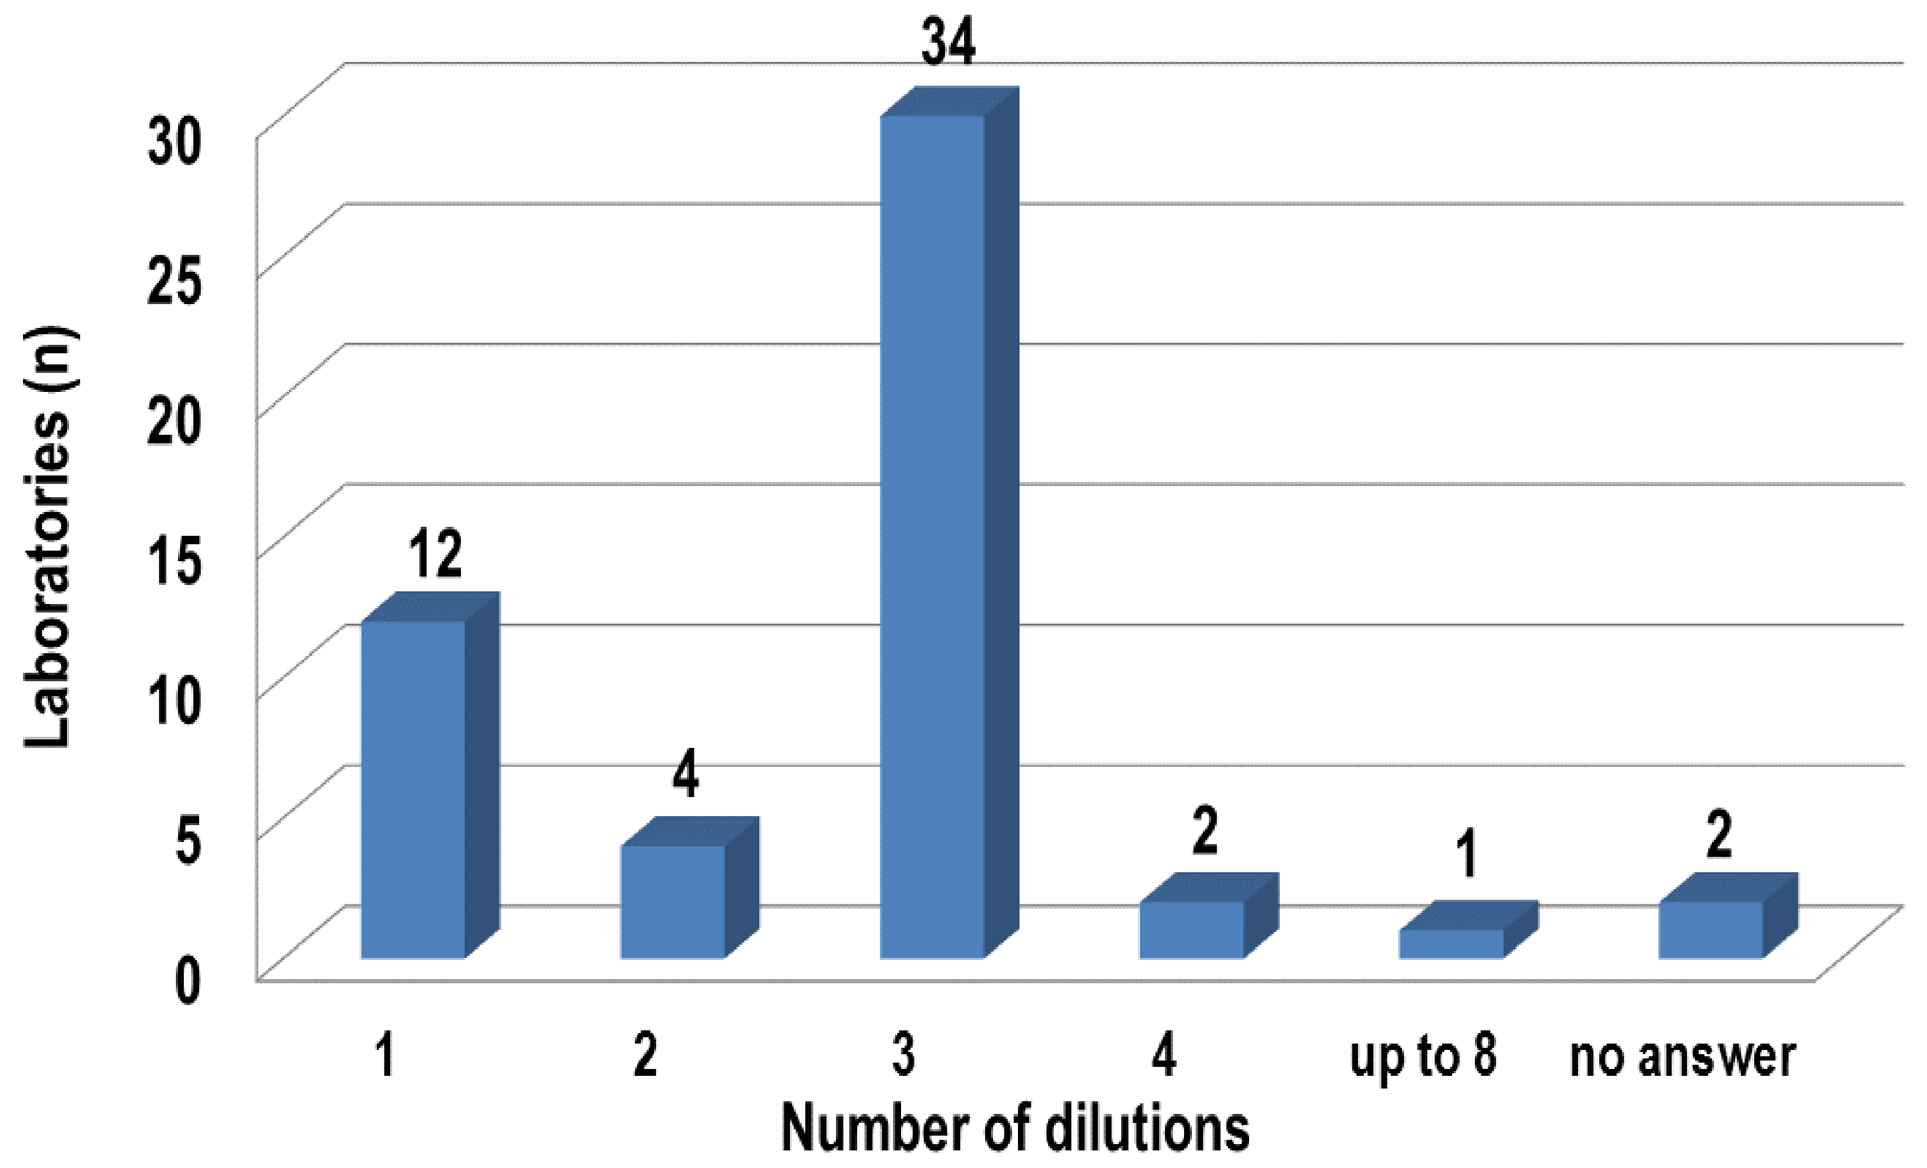
Laboratories (n)
0
5
10
15
20
25
30
12
4
34
2
1
2
1
2
3
4
up to 8
no answer
Number of dilutions

RESULTS – CHROMOGENIC ASSAYS

- Fifteen laboratories (15) have a chromogenic FVIII activity assay established, mainly in addition to a one-stage clotting method. Analytical instrument are from Instrumentation Laboratories (7), Siemens (5) or Sysmex (3). Both kinetic reading (9) and end point methods (5) are used, 1 laboratory provided no answer.

Figure 8: Analytical instruments

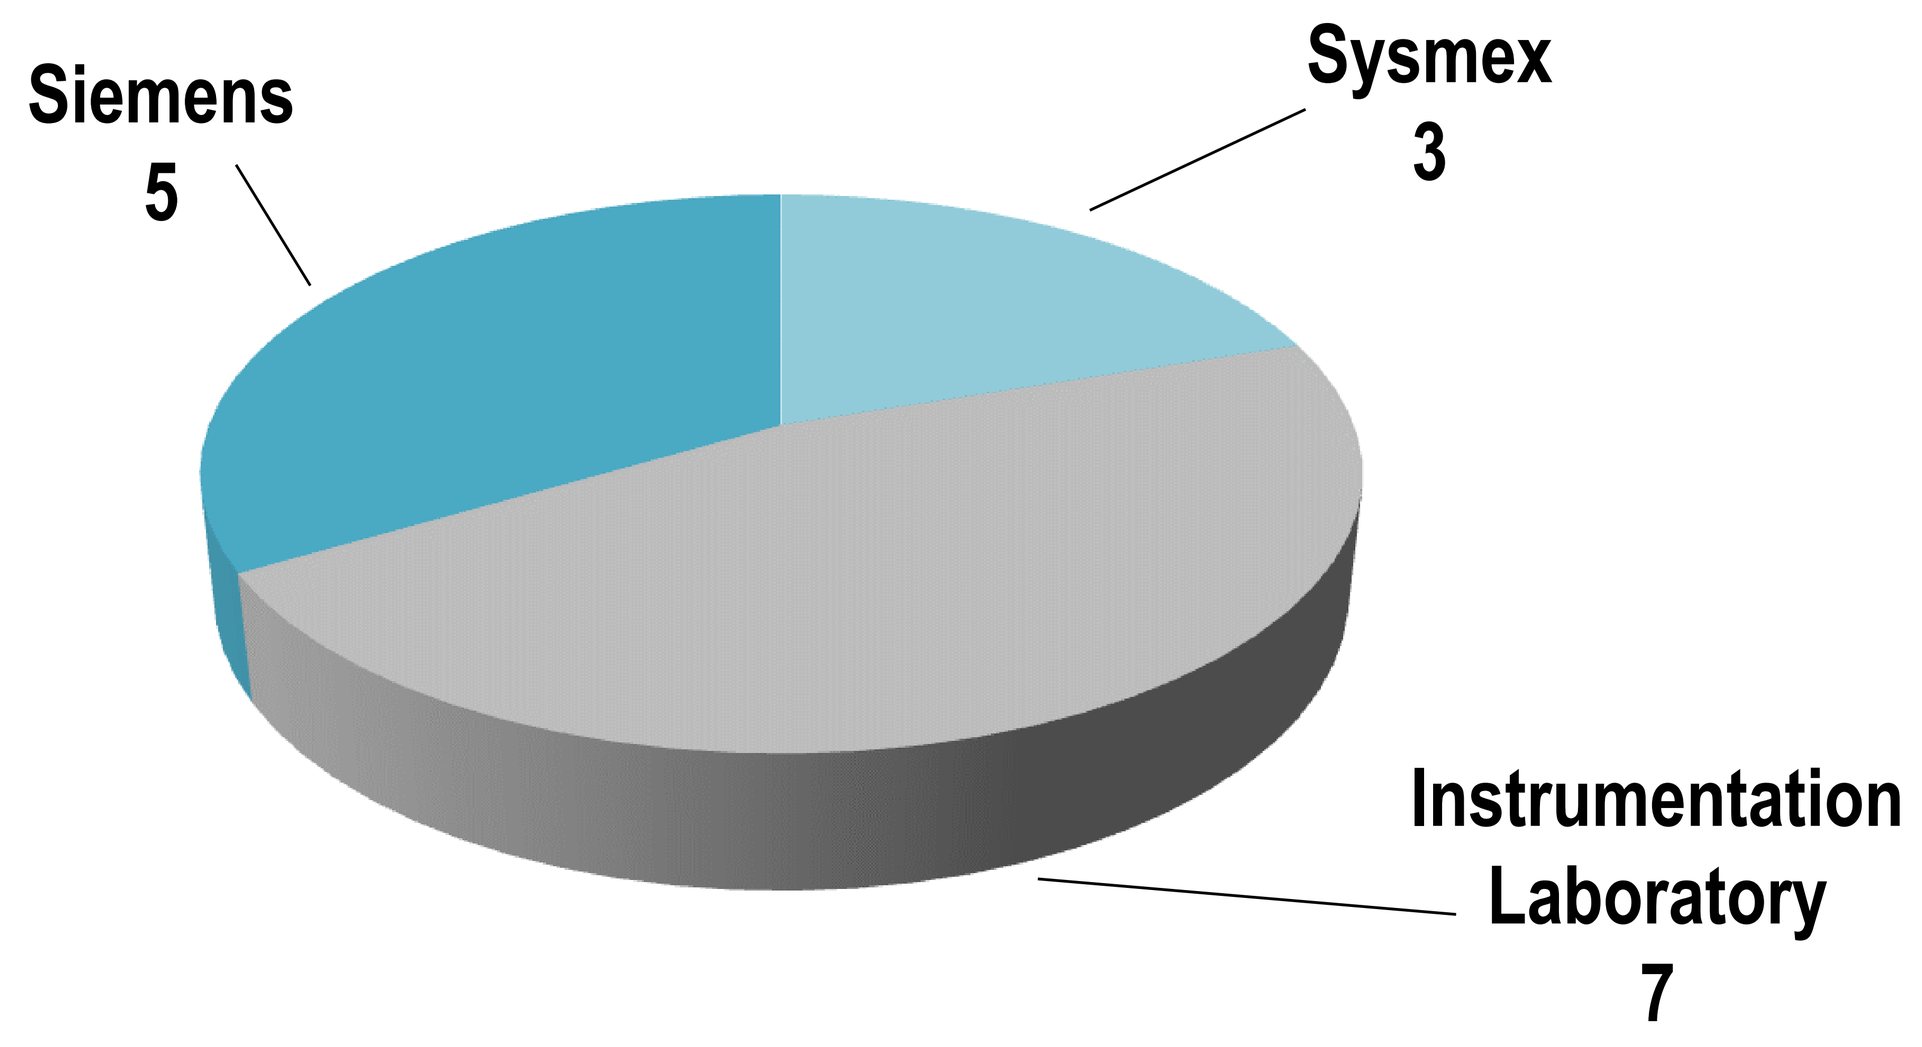
Siemens 5
Sysmex 3
Instrumentation Laboratory 7

Figure 9: Chromogenic assay reading mode

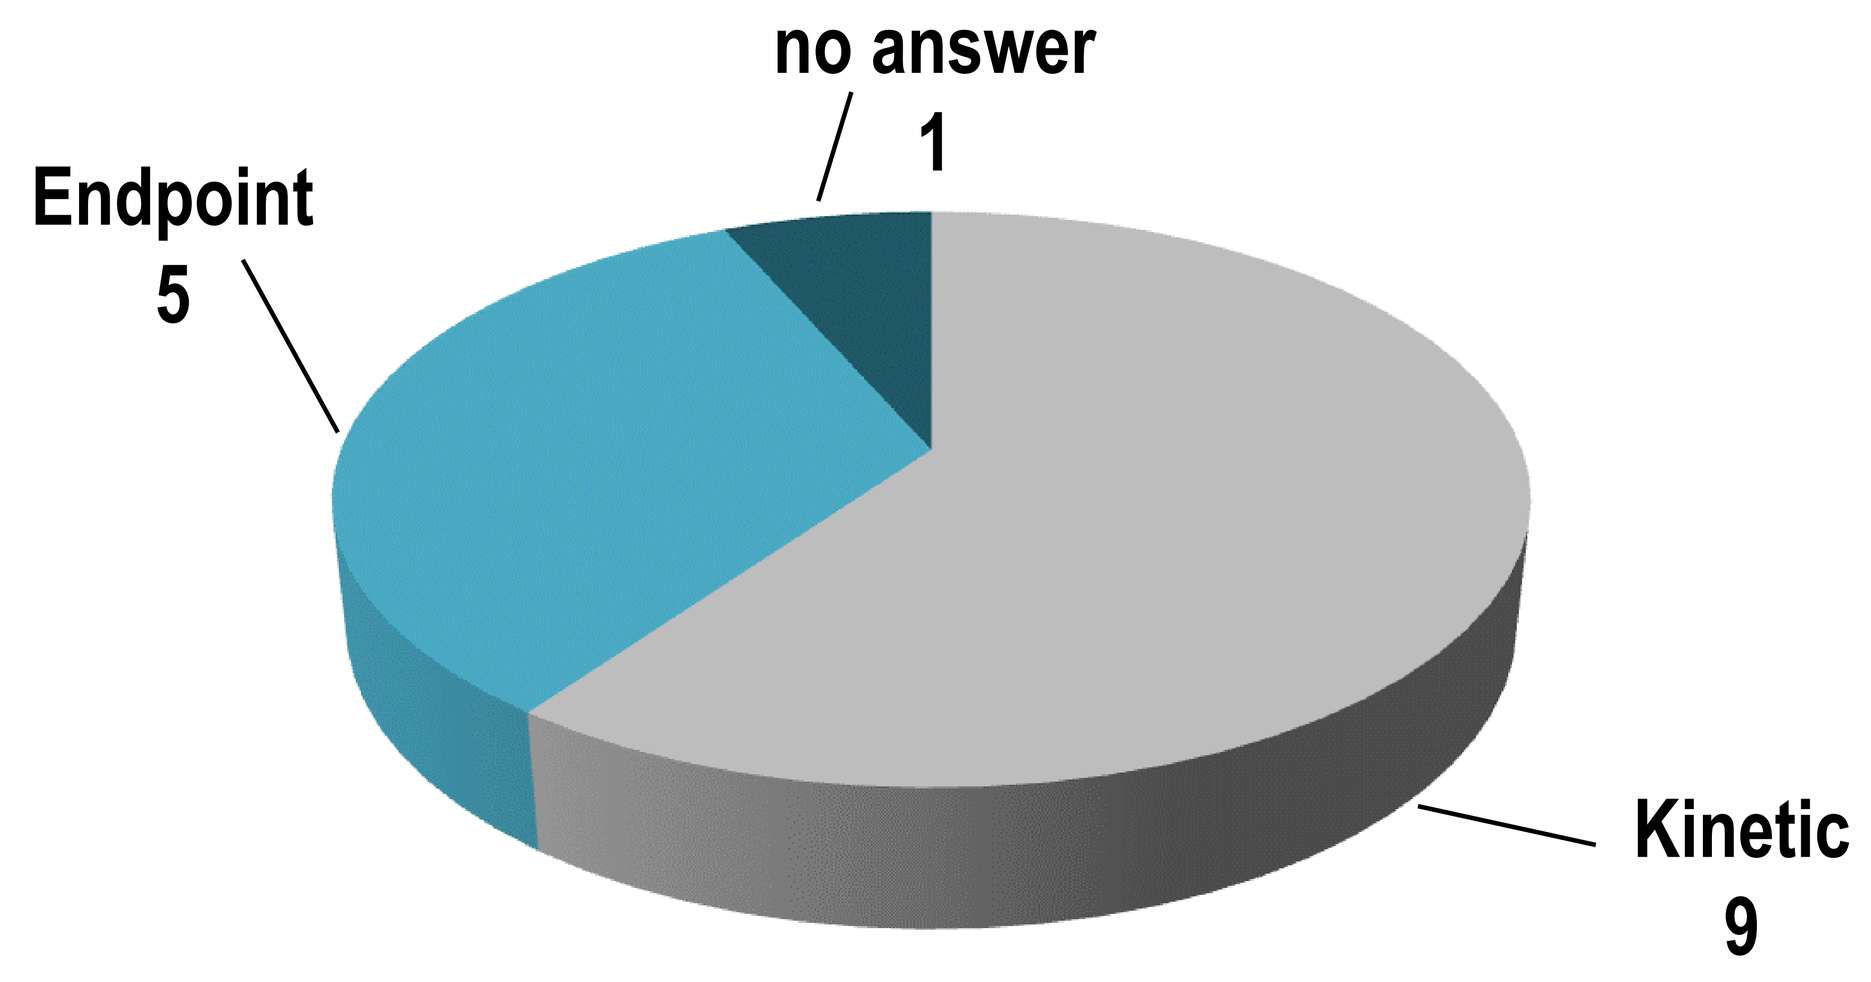
no answer 1
Endpoint 5
Kinetic 9

- Similarly to one-stage clotting assays, most laboratories are using stored reference curves, replaced either in regular intervals (4 laboratories) or with every change in critical reagent lots (5). Five (5) laboratories generate a new reference curve with every assay run. Reference material is mainly obtained from commercial sources (14), only one (1) laboratory is using an in-house normal plasma pool. Almost all laboratories (14) analyze only one single dilution of patient plasma, one (1) laboratory did not provide an answer.

CONCLUSION

- **The survey showed that in clinical practice currently the one-stage assay is the by far preferred type of FVIII assay**
 - **This confirmed previously published and reported expert opinion**
- **Most frequently silica based aPTT reagents are used followed by ellagic acid-based and kaolin reagents**
- **Reference plasma for assay calibration was mainly from commercial source, very often from the same manufacturer as the analytical instrument used**
- **Although a small portion of laboratories has also established a chromogenic FVIII activity assay only one laboratory was using this as the sole method to determine FVIII**

DISCLOSURES

Peter L. Turecek, Claudia Apostol and Herbert Gritsch are full-time employees of Baxalta Innovations GmbH, now part of Shire.

PosterSessionOnline.com
Poster Presented at:
DOI: 10.3252/pso.eu.WFH2016.2016
Laboratory issues
Peter L.
105--PP-W
WFH2016