Poster 96

Course of Target Joint Bleeding With Prophylaxis of a Pegylated Full-length Recombinant Factor VIII With Extended Half-life in Patients With Hemophilia A

Brigitt Abbuehl,[1] Werner Engl,[1] Lisa Patrone,[2] and Anne Prener[3*]

[1*]Shire, Vienna, Austria, [2*]Shire, Westlake Village, CA, and [3*]Shire, Cambridge, MA

INTRODUCTION

- Patients with severe hemophilia A have < 1% of normal factor VIII (FVIII) levels and experience frequent bleeding, which can be prevented by prophylaxis with FVIII replacement.[1]
- BAX 855, a polyethylene glycol (peg)ylated, full-length, recombinant FVIII (rFVIII), is built on the plasma/albumin-free manufacturing platform of rAHF-PFM (ADVATE). Controlled pegylation was chosen to extend the FVIII half-life while maintaining the integrity of the ADVATE protein.[2]
- A first-in-human phase 1 clinical study with BAX 855 demonstrated that the half-life in the circulation is extended by up to 1.5 times compared to ADVATE and that single infusions were well tolerated.[3]
- The BAX 855 pivotal study confirmed the extended half-life of BAX 855 and demonstrated the efficacy and safety of BAX 855 for prophylaxis and for the treatment of bleeding in previously treated adolescents and adults with severe hemophilia A.[4]
- The BAX 855 pediatric study demonstrated the safety and efficacy of BAX 855 for prophylaxis and for the treatment of bleeding episodes in previously children with severe hemophilia A which included confirmation of its extended half-life.[5]
- The BAX 855 surgery study demonstrated the safety and efficacy of BAX 855 for perioperative management.[6]
- Patients from these studies could continue treatment in the continuation study.
- In an integrated analysis from these studies, long-term BAX 855 twice weekly prophylaxis was shown to be efficacious in patients with hemophilia A.[7]

OBJECTIVE

- Data from the BAX 855 clinical program (3 completed and 2 ongoing studies) were integrated to evaluate efficacy. To evaluate the course of target joints, 3 of the studies were included in the analysis; pivotal, pediatric, and continuation (refer to Table 1).

METHODS

Table 1: BAX 855 Studies

Description	IDs for ClinicalTrials.gov clinicaltrialsregister.eu	Study Status	Planned Treatment
Phase 1: Pharmacokinetics (PK), single infusion tolerability	NCT01599819	Complete	PK: 30 or 60 ± 5 IU IU/kg
***Pivotal**: Phase 2/3, PK, efficacy, safety, immunogenicity	NCT01736475 EudraCT 2012-003599-38	Complete	PK: 45 ± 5 IU/kg Prophylaxis: 45 ± 5 IU/kg twice weekly On-demand treatment: 10 to 60 ± 5 IU/kg
***Pediatric**: Phase 3, PK, efficacy, safety, immunogenicity	NCT02210091 EudraCT 2014-000742-30	Complete	PK: 60 ± 5 IU/kg Prophylaxis: 50 ± 10 IU/kg twice weekly
Surgery: Phase 3, perioperative hemostasis, safety	NCT01913405 EudraCT 2013-001359-11	Ongoing	Tailored dose to achieve FVIII target levels for: Major procedures: 80 to 100% of normal Minor procedures: 30 to 60% of normal
***Continuation**: Phase 3b, long-term efficacy, safety, immunogenicity	NCT01945593 EudraCT 2014-005477-37	Ongoing	Prophylaxis: ≤ 12 years: 45 ± 5 IU/kg < 12 years: 50 ± 10 IU/kg (may adjust to 45-80 IU/kg) twice weekly** or PK-tailored, targeting ≥3% FVIII trough levels

*Studies included for the evaluation of the course of target joints during twice weekly prophylaxis.
**May have been reduced based on spontaneous bleeding in the prior treatment period.

WFH World Congress; July 24–28, 2016; Orlando, USA

METHODS continued

Figure 1: Patient Disposition – 234 Treated Patients, 215 on Prophylaxis

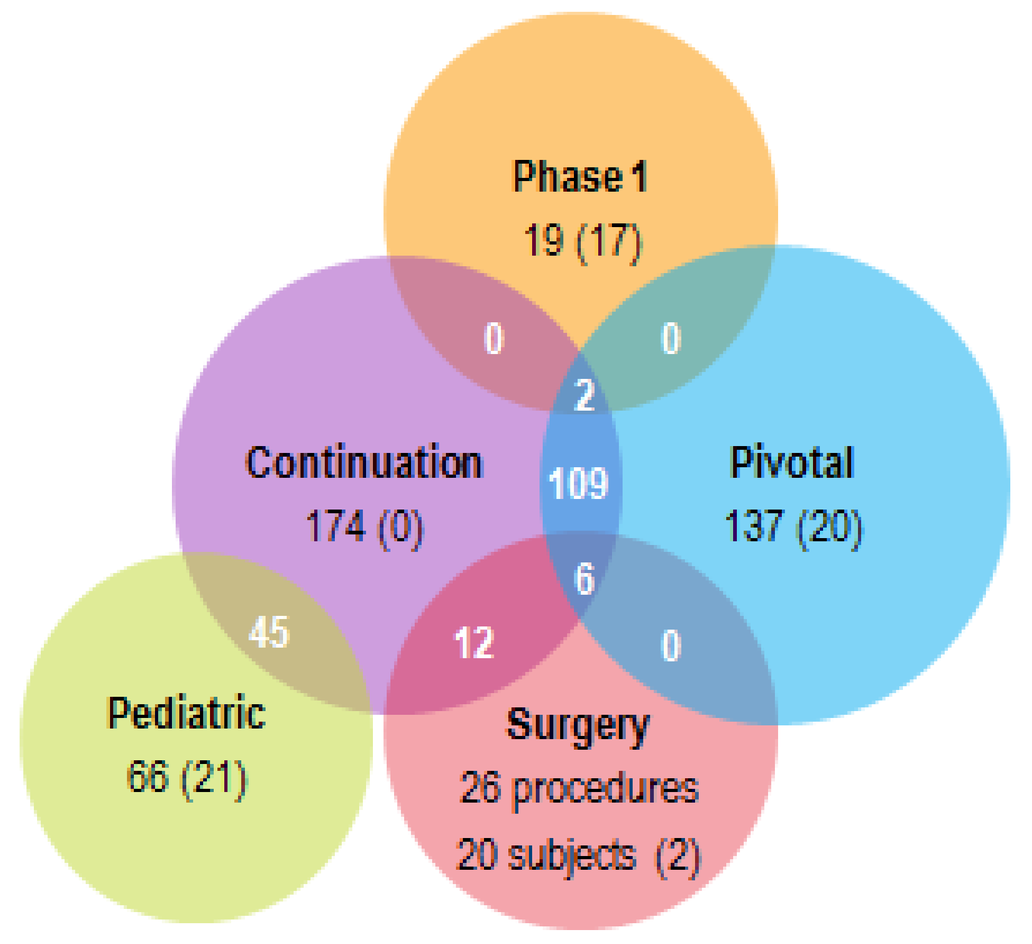
Phase 1
19 (17)
Continuation
174 (0)
Pivotal
137 (20)
Pediatric
66 (21)
Surgery
26 procedures
20 subjects (2)
0
0
2
109
6
45
12
0

- N (n): Number of patients treated in the study (number of patients unique to that study); others were treated in 1 or more studies
- Data from the pivotal, pediatric, and continuation studies were evaluated in the course of target joints analysis

Prophylactic Treatment Regimens

- All patients initially received twice weekly fixed dose prophylaxis for approximately 6 months as follows:
 - 45 ± 5 IU/kg for adolescents and adults ≥ 12 years of age
 - 50 ± 10 IU/kg for children < 12 years of age
- After the initial period, adolescents and adults with low spontaneous annualized bleeding rates (sABRs) may have reduced their dosing frequency for the next 6-month period; a further reduction was possible for patients who maintained low sABRs. Patients with higher sABRs may have resumed twice weekly prophylaxis at a higher dose (45–80 IU/kg).[7] Thus, the subsequent 6-month periods of twice weekly prophylaxis are likely to include more patients with higher bleeding rates. As of the data cut-off date for this analysis, 68 patients were on a reduced dosing frequency regimen in the continuation study.
- Alternatively, after the initial period, patients may have been treated on PK-tailored prophylaxis targeting ≥ 3% FVIII trough levels. As of the data cut-off date for this analysis, 15 patients were on a PK-tailored regimen in the continuation study.

RESULTS

Demographics

- **Age**: The mean ±SD (minimum to maximum) was 23.6 ±15.4 (1 to 60) years of age.
- **Gender**: Of 234 treated patients, 1 was female and the remaining were male.
- **Race**: Most (74.4%) treated patients were White, 21.8% were Asian, 2.6% were Black, and the remaining 0.9% were of other race(s).

Figure 2: Baseline Target Joints and Arthropathy Status (N=203)

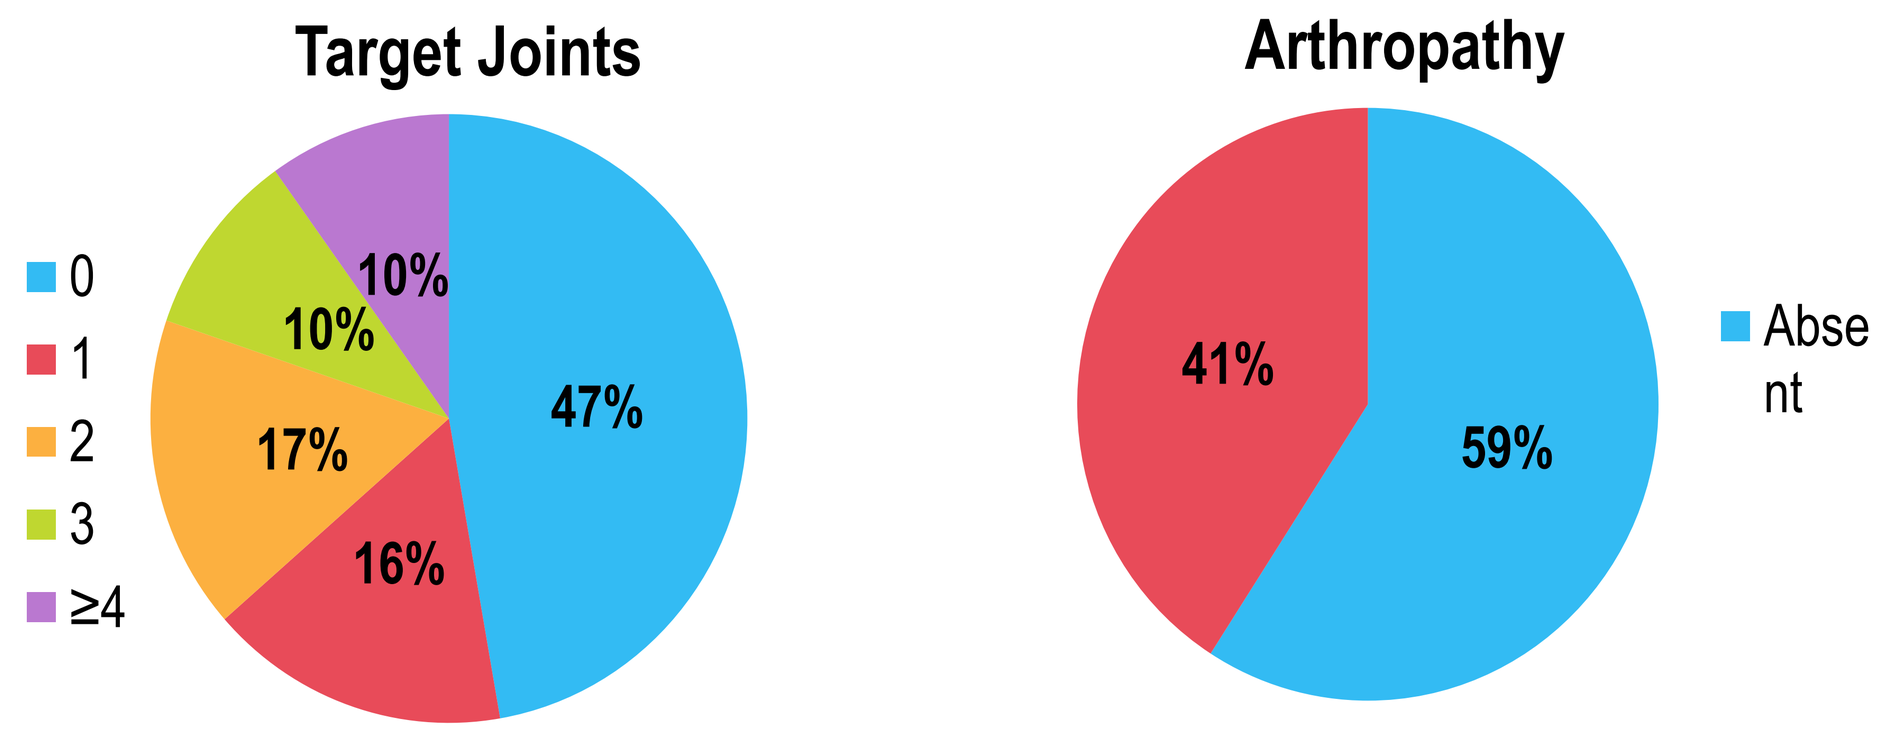
Target Joints
0
1
2
3
≥4
47%
16%
17%
10%
10%
Arthropathy
41%
59%
Abse
nt

	Patients	NO Target Joints	NO Arthropathy
Adults (≥ 18 years)	112	27%	32%
Adolescents (12 to < 18 years)	25	56%	84%
Older children (6 to < 12 years)	34	68%	91%
Younger children (< 6 years)	32	91%	100%

- A Target joint was defined as a single joint (ankle, knee, hip or elbow) with ≥ 3 spontaneous bleeding episodes in any consecutive 6-month period
- Arthropathy status was reported at screening of the initial study of participation

RESULTS

Figure 3: Percent of Patients with Joint Bleeding Episodes Over Time on Twice Weekly Prophylaxis

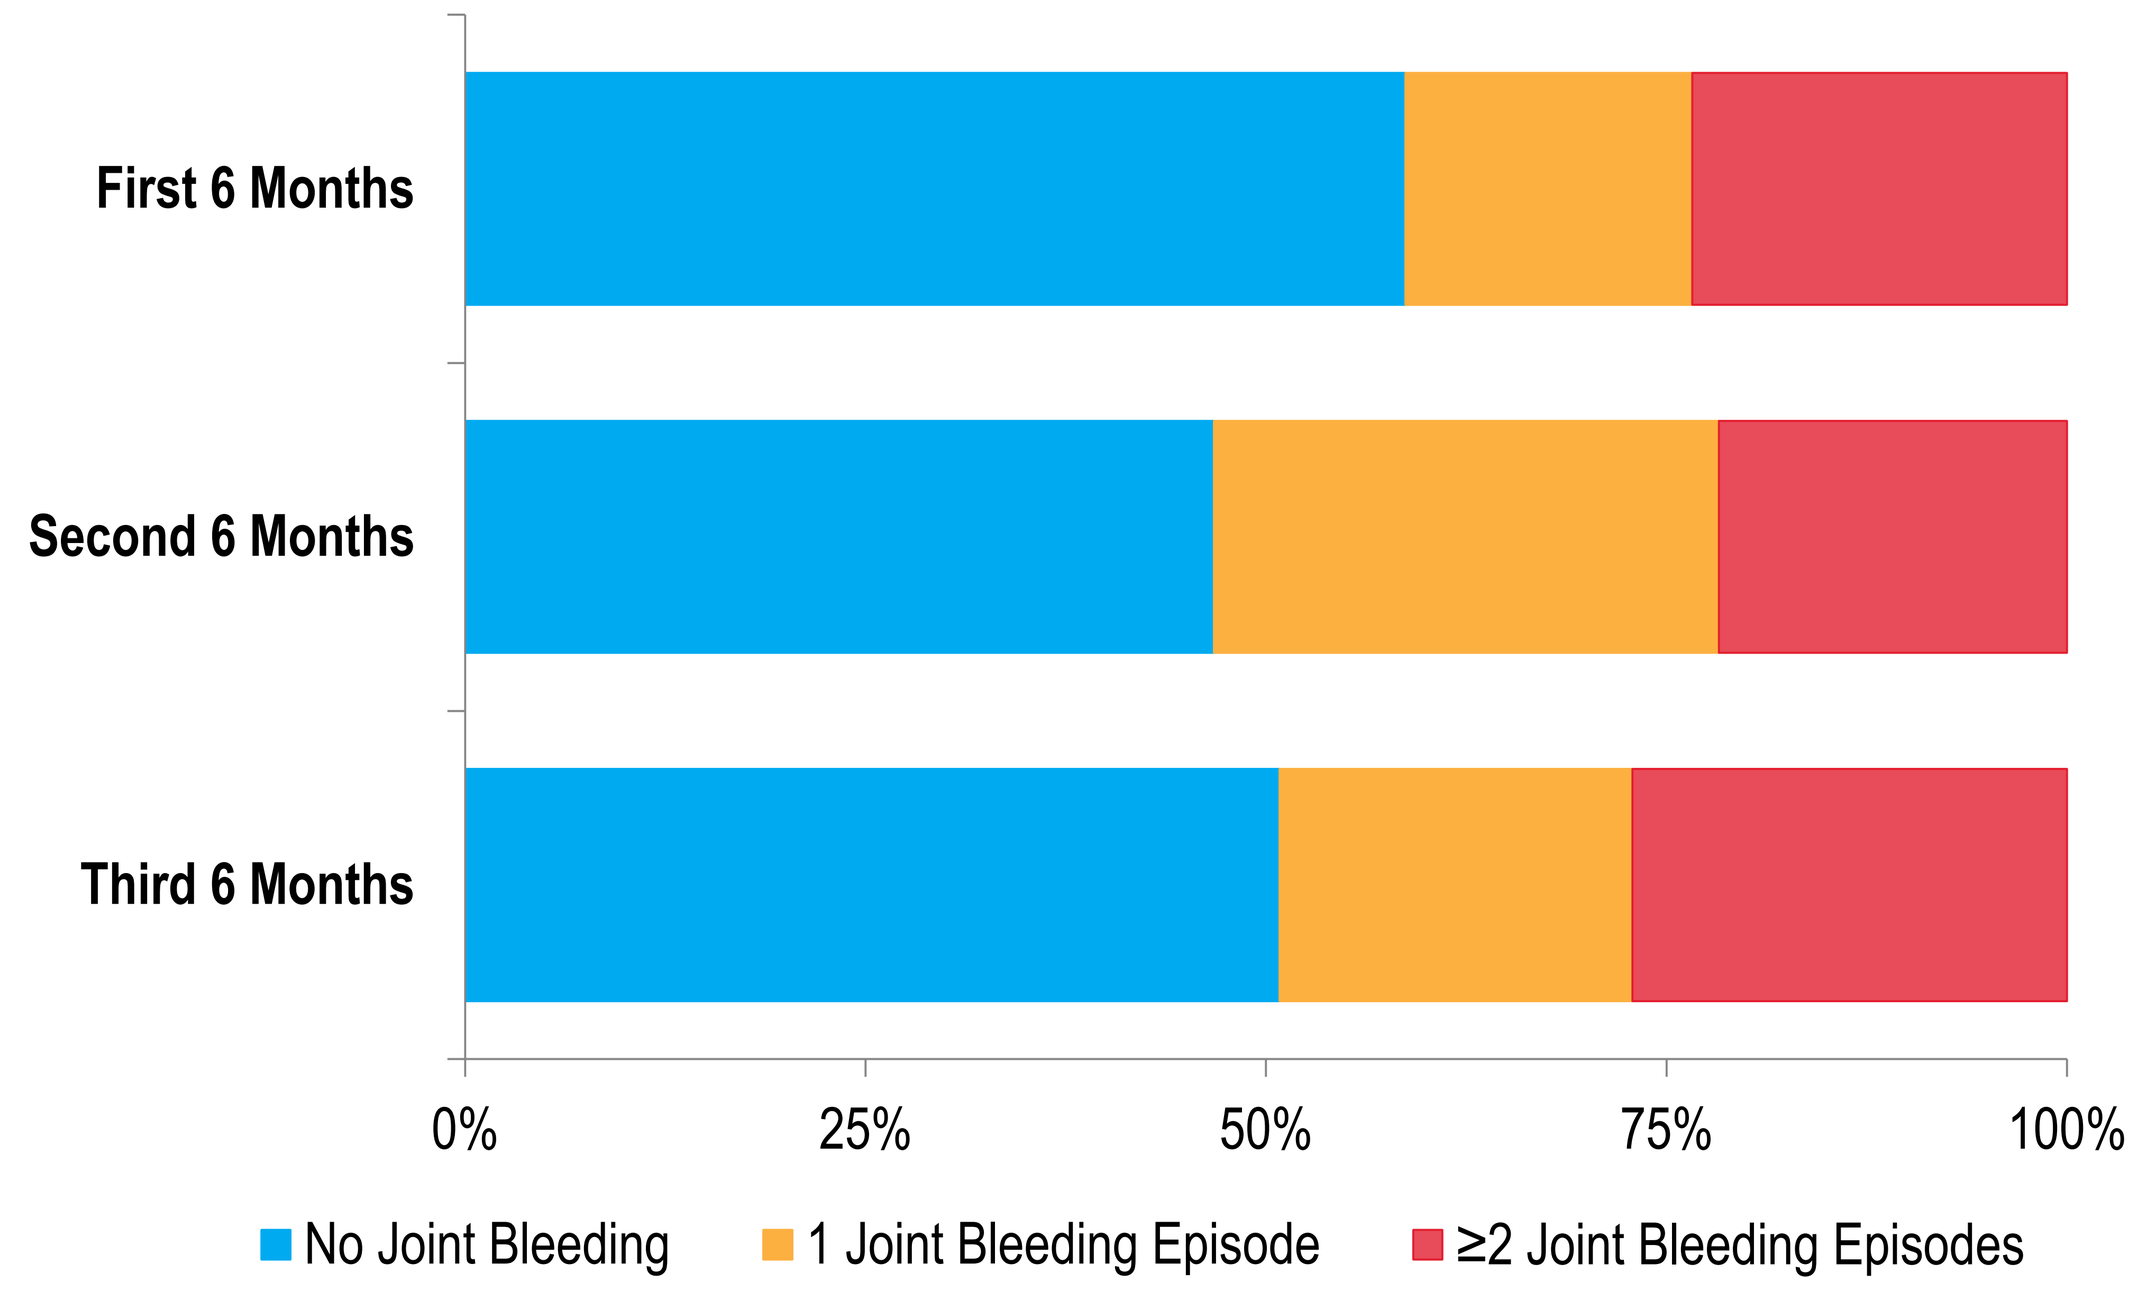
First 6 Months
Second 6 Months
Third 6 Months
0%
25%
50%
75%
100%
No Joint Bleeding
1 Joint Bleeding Episode
≥2 Joint Bleeding Episodes

- Age Groups included:
 - First 6 Months, n = 201: 63 Children (2 were 1 month to < 2 years; 30 were 2 to < 6 years; 31 were 6 to < 12 years), 24 Adolescents, and 114 Adults
 - Second 6 Months, n = 92: 14 Adolescents and 78 Adults
 - Third 6 Months, n = 59: 11 Adolescents and 48 Adults
- 59% of patients had no joint bleeding episodes during the first 6 months of twice weekly prophylaxis with BAX 855.
- Fewer (47% and 51%) patients had no joint bleeding episodes over consecutive treatment periods, which were without the children (who had fewer target joints and less arthropathy at screening).

Figure 4: Percent of Patients with Target Joint (TJ) Bleeding Over Time on Twice Weekly Prophylaxis

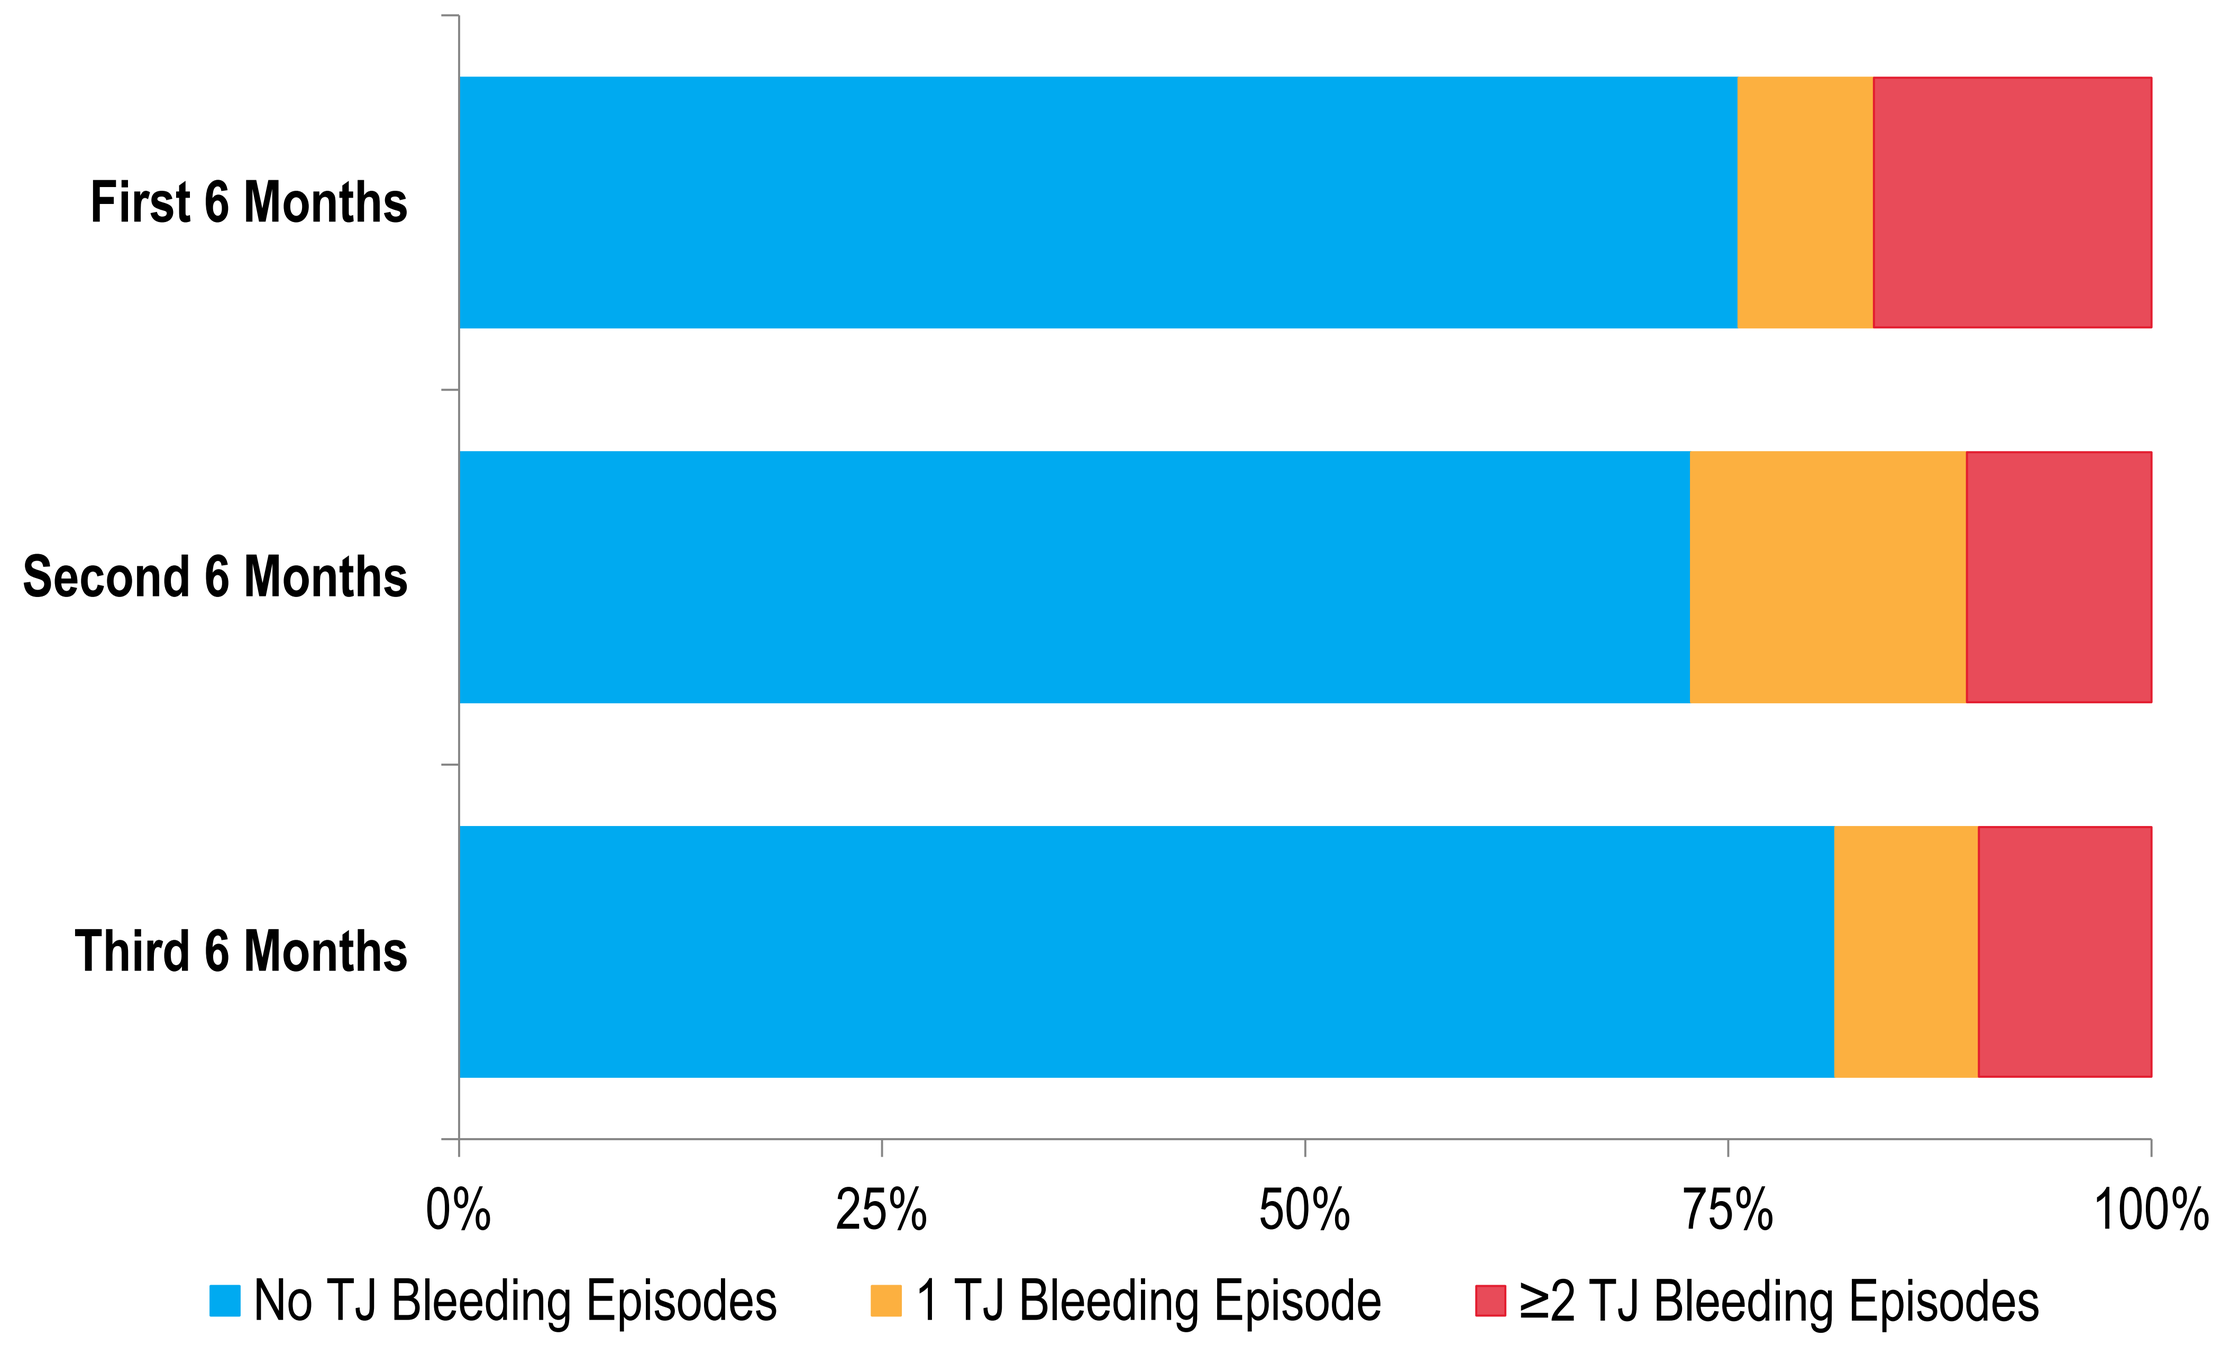
First 6 Months
Second 6 Months
Third 6 Months
0%
25%
50%
75%
100%
No TJ Bleeding Episodes
1 TJ Bleeding Episode
≥2 TJ Bleeding Episodes

- For the numbers of patients and age groups included, refer to Figure 3.
- 76% of patients had no target joint bleeding during the first 6 months of twice weekly prophylaxis with BAX 855.
- The percent of patients with no target joint bleeding was similar over consecutive 6-month treatment periods, which were without the children (who had fewer target joints and less arthropathy at screening).

ACKNOWLEDGEMENTS AND DISCLOSURES

The authors recognize with gratitude the investigators and patients who participated in the studies. The studies were sponsored by Baxalta US Inc. and Baxalta Innovations GmbH; Baxalta, now part of Shire. If you have any additional questions, please feel free to contact our Medical Information at medinfo@baxalta.com.

*Author an employee of Baxalta ([1]Baxalta Innovations GmbH,[2,3]Baxalta US Inc), now part of Shire.

Figure 5: Percent of Patients by Age Group with Target Joint (TJ) Bleeding Over Time on Twice Weekly Prophylaxis

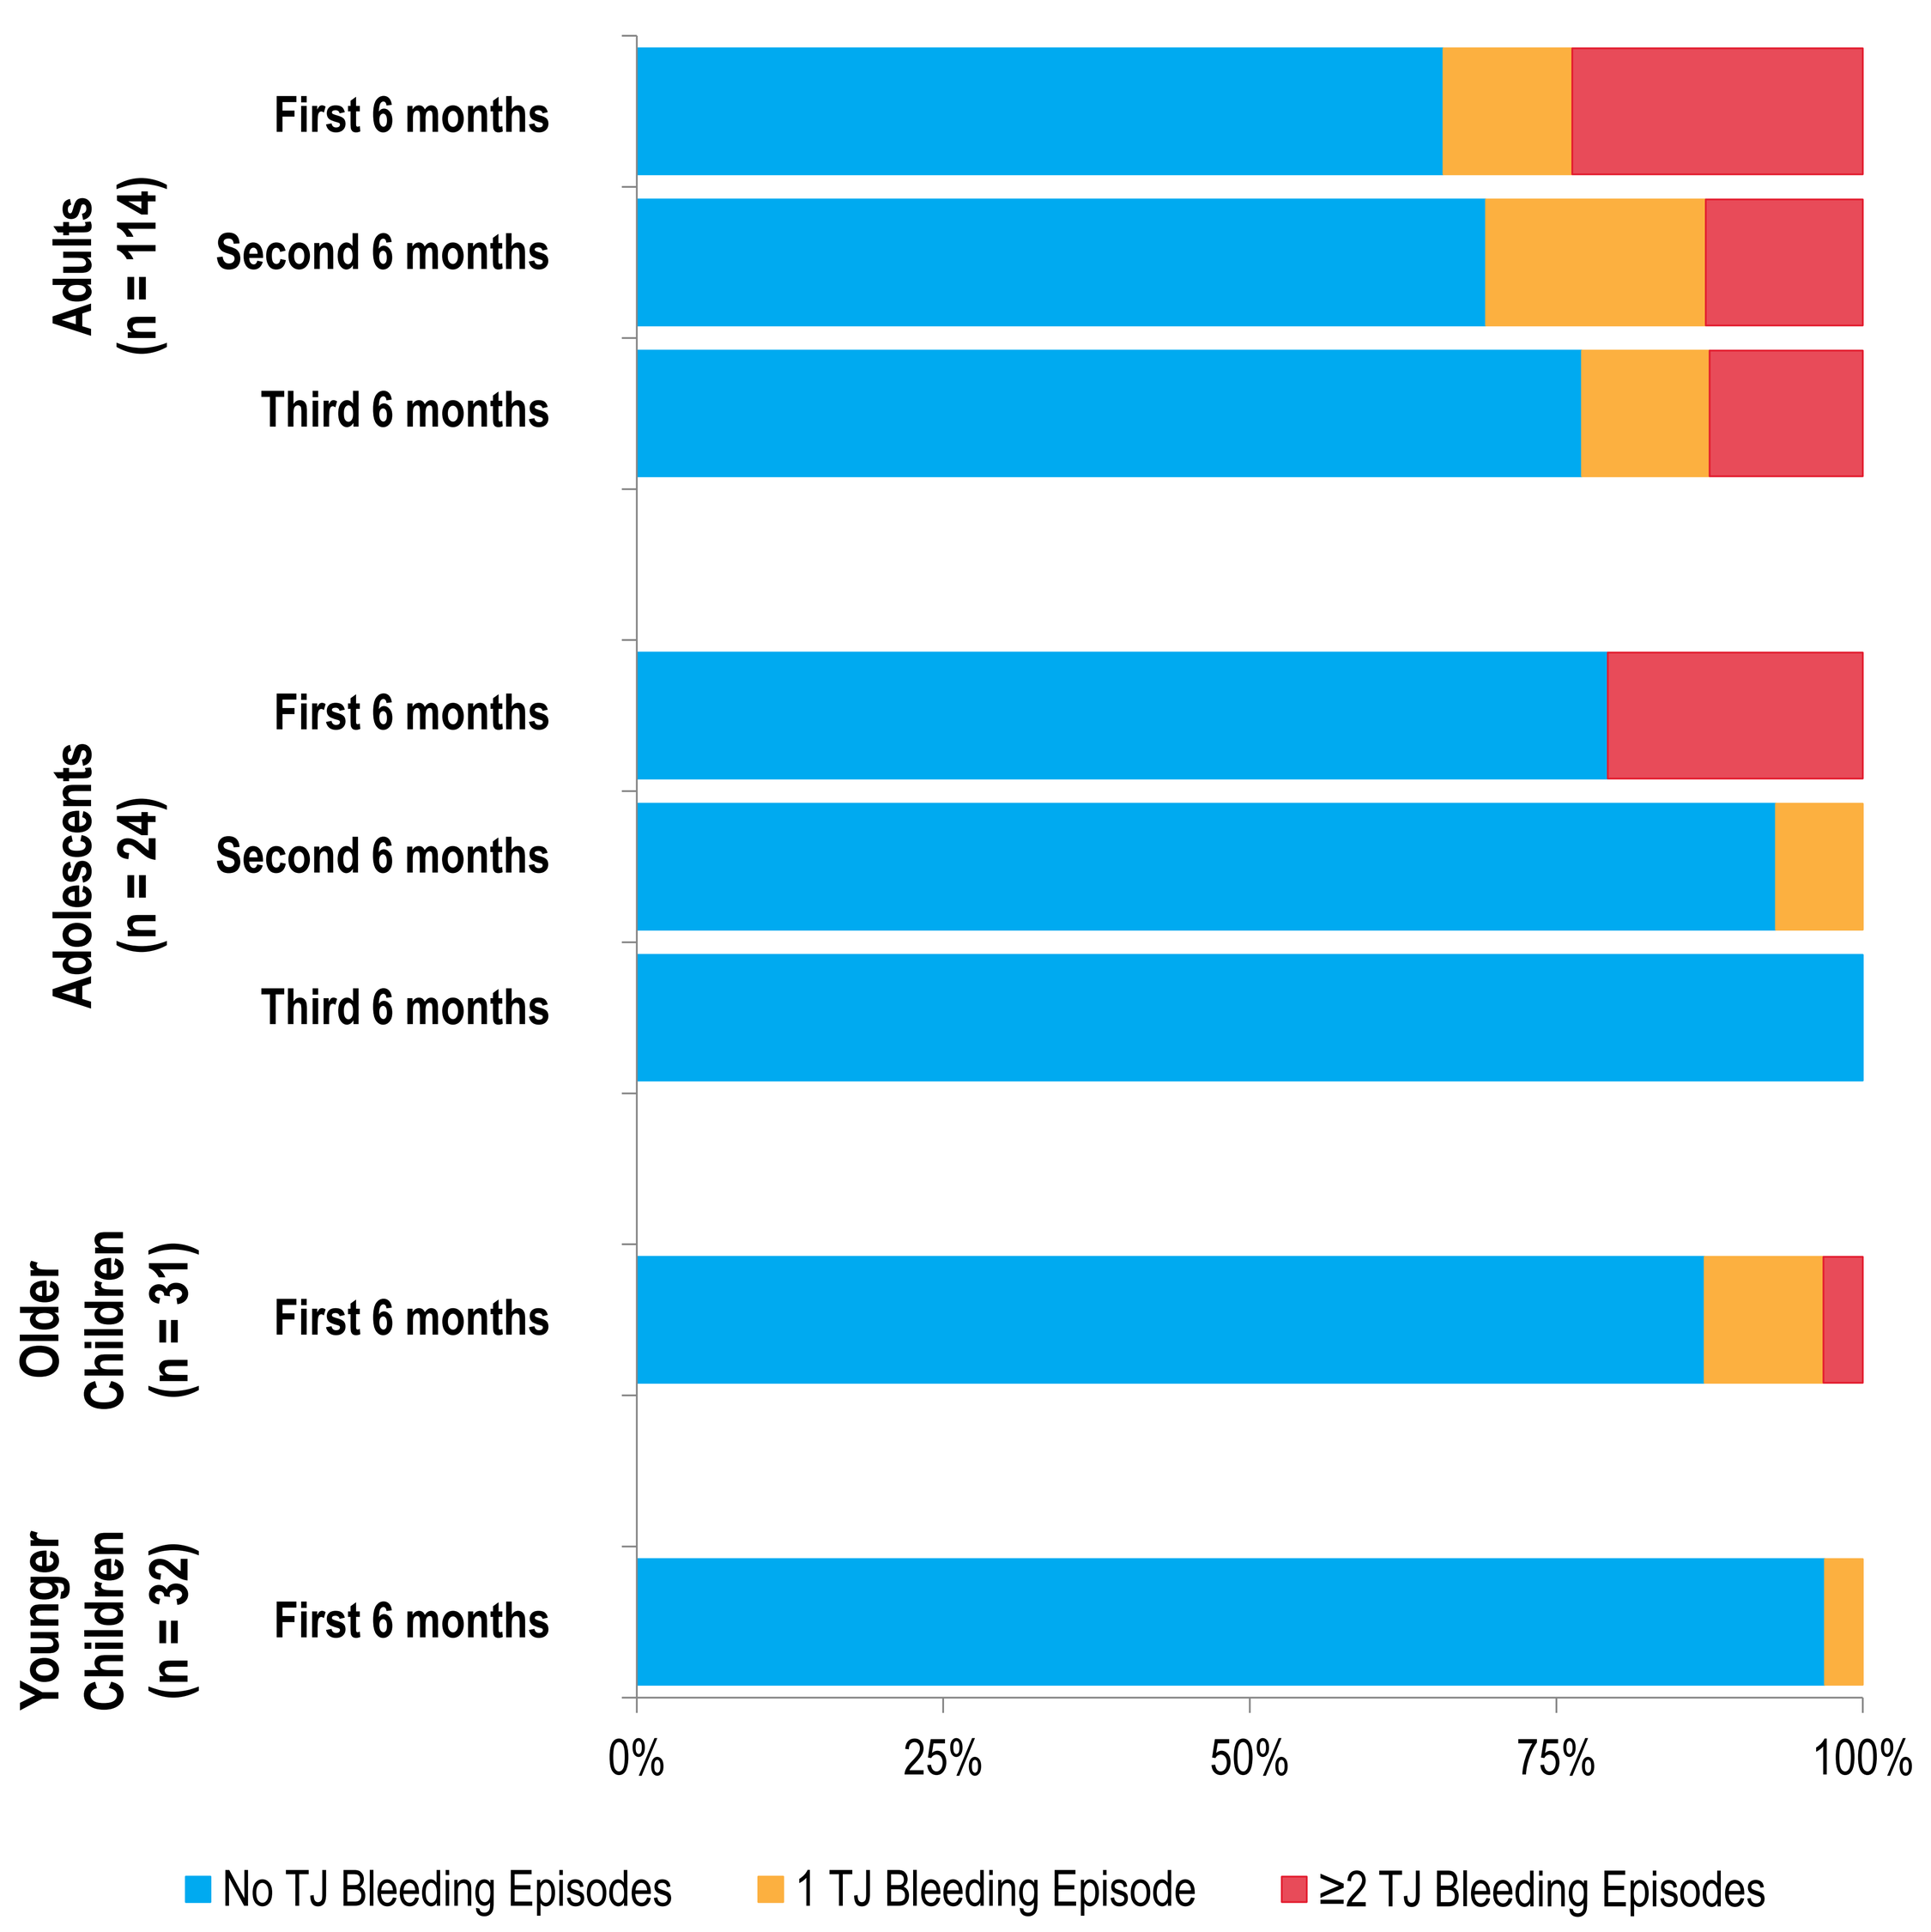
Adults
(n = 114)
First 6 months
Second 6 months
Third 6 months
Adolescents
(n = 24)
First 6 months
Second 6 months
Third 6 months
Older
Children
(n = 31)
First 6 months
Younger
Children
(n = 32)
First 6 months
0%
25%
50%
75%
100%
No TJ Bleeding Episodes
1 TJ Bleeding Episode
≥2 TJ Bleeding Episodes

- None of the children completed a full second (or third 6-month) treatment period by the time of the data cut-off for this analysis.
- The percent of **adults** on twice weekly prophylaxis with no target joint bleeding was maintained (or increased slightly) over consecutive treatment periods.
- The percent of **adolescents** on twice weekly prophylaxis with no target joint bleeding increased over consecutive treatment periods (ie from 80% in the first period to 100% in the third period).
- During the first 6-month treatment period, over half of the target joints reverted to non-target joints (15 of 23; data not shown in the figure).

CONCLUSION

- **These results demonstrate that during continuous twice weekly prophylaxis with BAX 855 of up to 18 months, target joint bleeding remains low or even disappears, as shown in the group of adolescents.**

REFERENCES

1. Srivastava A, et al. Guidelines for the management of hemophilia. Haemophilia. 2013;19:e1-e47.
2. Turecek PL, et al. BAX 855, a PEGylated rFVIII product with prolonged half-life. *Hämostaseologie*. 2012;32(1): S29-S38.
3. Bevan D, et al. A Phase 1 study of safety and pharmacokinetics (PK) of BAX 855, a longer acting pegylated full-length recombinant Factor VIII (PEG-rFVIII), in patients (pts) with severe haemophilia A. *Haemophilia.* 2013;(s2):Po053, pp.32.
4. Konkle BA, et al. Pegylated, full-length, recombinant factor VIII for prophylactic and on-demand treatment of severe hemophilia A. *Blood.* 2015;126(9):1078-1085.
5. Mullins E, et al. Safety and efficacy of a pegylated full-length recombinant factor VIII with extended half-life in previously treated children with hemophilia A. 2016 WFH abstract 622.
6. Brand B, et al. Efficacy and safety of pegylated full-length recombinant factor VIII with extended half-life for perioperative hemostasis in hemophilia A patients. *Haemophilia* (manuscript in press).
7. Abbuehl B, et al. Evaluation of long-term prophylaxis with a pegylated full-length recombinant factor VIII with extended half-life in patients with hemophilia A. 2016 WFH abstract 864.

PosterSessionOnline.com

Poster Presented at:

WFH 2016 WORLD CONGRESS Orlando, USA July 24-28

DOI: 10.3252/pso.eu.WFH2016.2016

Hemophilia - clinical
Lisa Patrone

96--PP-T

WFH2016