Effect of factor VIII coagulation activity (FVIII:C) on the risk of spontaneous bleeding following treatment with rFVIII (turoctocog alfa) in patients with severe hemophilia A

P-T-91

A Velsing Groth;[1] H Agersø;[1] I Matytsina;[1] N Tripkovic;[2] R Viig Overgaard[1]
[1]Novo Nordisk A/S, Søborg, Denmark; [2]Novo Nordisk Health Care, Zurich, Switzerland

Objective

- To assess the relationship between FVIII:C and annualized bleeding rate (ABR) in patients with severe hemophilia A after treatment with turoctocog alfa in the guardian™1 and 3 (pivotal phase) and guardian™2 extension trials.

Conclusions

- In patients with severe hemophilia A treated with turoctocog alfa, ABR was lower at times of higher FVIII activity, indicating an activity–response relationship.
- This activity–response relationship was evident in both the pivotal and extension phases, with lower ABRs in the extension phase.
- The relationship was also apparent for adults and children, although children had lower ABRs than adults.

Introduction

- In phase 3 trials, turoctocog alfa – a B-domain-truncated recombinant factor VIII (rFVIII) product – demonstrated favorable safety and efficacy in adults and adolescents (guardian™1) and children (guardian™3) with severe hemophilia A.[1,2]
- guardian™2 is a large extension trial that included patients from guardian™1 and guardian™3.[3]
- Data from these trials were used to determine the relationship between FVIII:C, assumed proportional to turoctocog alfa concentration, and ABR for spontaneous bleeds after treatment with turoctocog alfa.

Methods

Patients and trial design

- guardian™1 and guardian™3 were multinational phase 3 trials assessing prophylaxis with turoctocog alfa in adult/adolescent (aged 12–65 years) and pediatric (aged 0–11 years) patients, respectively.[1,2]
- All patients had severe hemophilia A (FVIII ≤1%) without inhibitors and ≥150 (adults/adolescents) or ≥50 (children) exposure days to FVIII.
- Patients completing guardian™1 or 3 could continue into the open-label guardian™2 extension trial.[3]

Predicting FVIII:C profiles

- guardian™1 and 3 included single-dose pharmacokinetic (PK) assessment after the first dose and after 3 months' treatment, which involved 9 (adults/adolescents) or 6 (children) sampling time points up to 48 hours post infusion.[1,2]
- A population PK model derived from these trials was applied to diary-recorded dosing data from all patients with appropriate records to obtain predicted PK profiles over the entire trial duration for each patient. Each patient's time in the trial was divided into different FVIII:C categories.

Estimating the relationship between FVIII:C and ABR

- Bleeding episodes were also recorded in patient diaries. To analyze the relationship between FVIII:C and ABR, FVIII:C was categorized into five clinically meaningful groups: 0–1%, >1–5%, >5–20%, >20–50%, and >50%.
- ABRs were estimated for each FVIII:C category using negative binomial regression and predictions of a parametric model.
- Relationships between ABR and mean FVIII:C were evaluated for:
 - Overall dataset: guardian™1 and 3 ('pivotal phase').
 - Trial phase: pivotal versus extension phase.
 - Age: adults (aged ≥12 years) versus children (aged 0–<12 years).

Results

Patients and turoctocog alfa exposure

- The number of patients and patient years of exposure (PYE) in the analyses populations were:
 - Pivotal phase (n=212; PYE=100.18) versus extension phase (n=187; PYE=237.78).
 - Adults (n=281; PYE=260.53) versus children (n=118; PYE=77.43).

Exposure–response

- ABR showed exposure–response versus predicted FVIII:C following turoctocog alfa administration (Table 1).

ABR by trial phase and patient age

- ABRs were lower in the extension versus the pivotal phase (Figure 1A), and lower in children versus adult patients (Figure 1B).

Figure 1 Negative binomial estimates of ABR (mean and 95% CI) for each FVIII:C category investigated (0–1%, >1–5%, >5–20%, >20–50%, and >50%) along with predictions of a parametric E_{max} model splitting data by trial phase (A) and by patient age group (B).

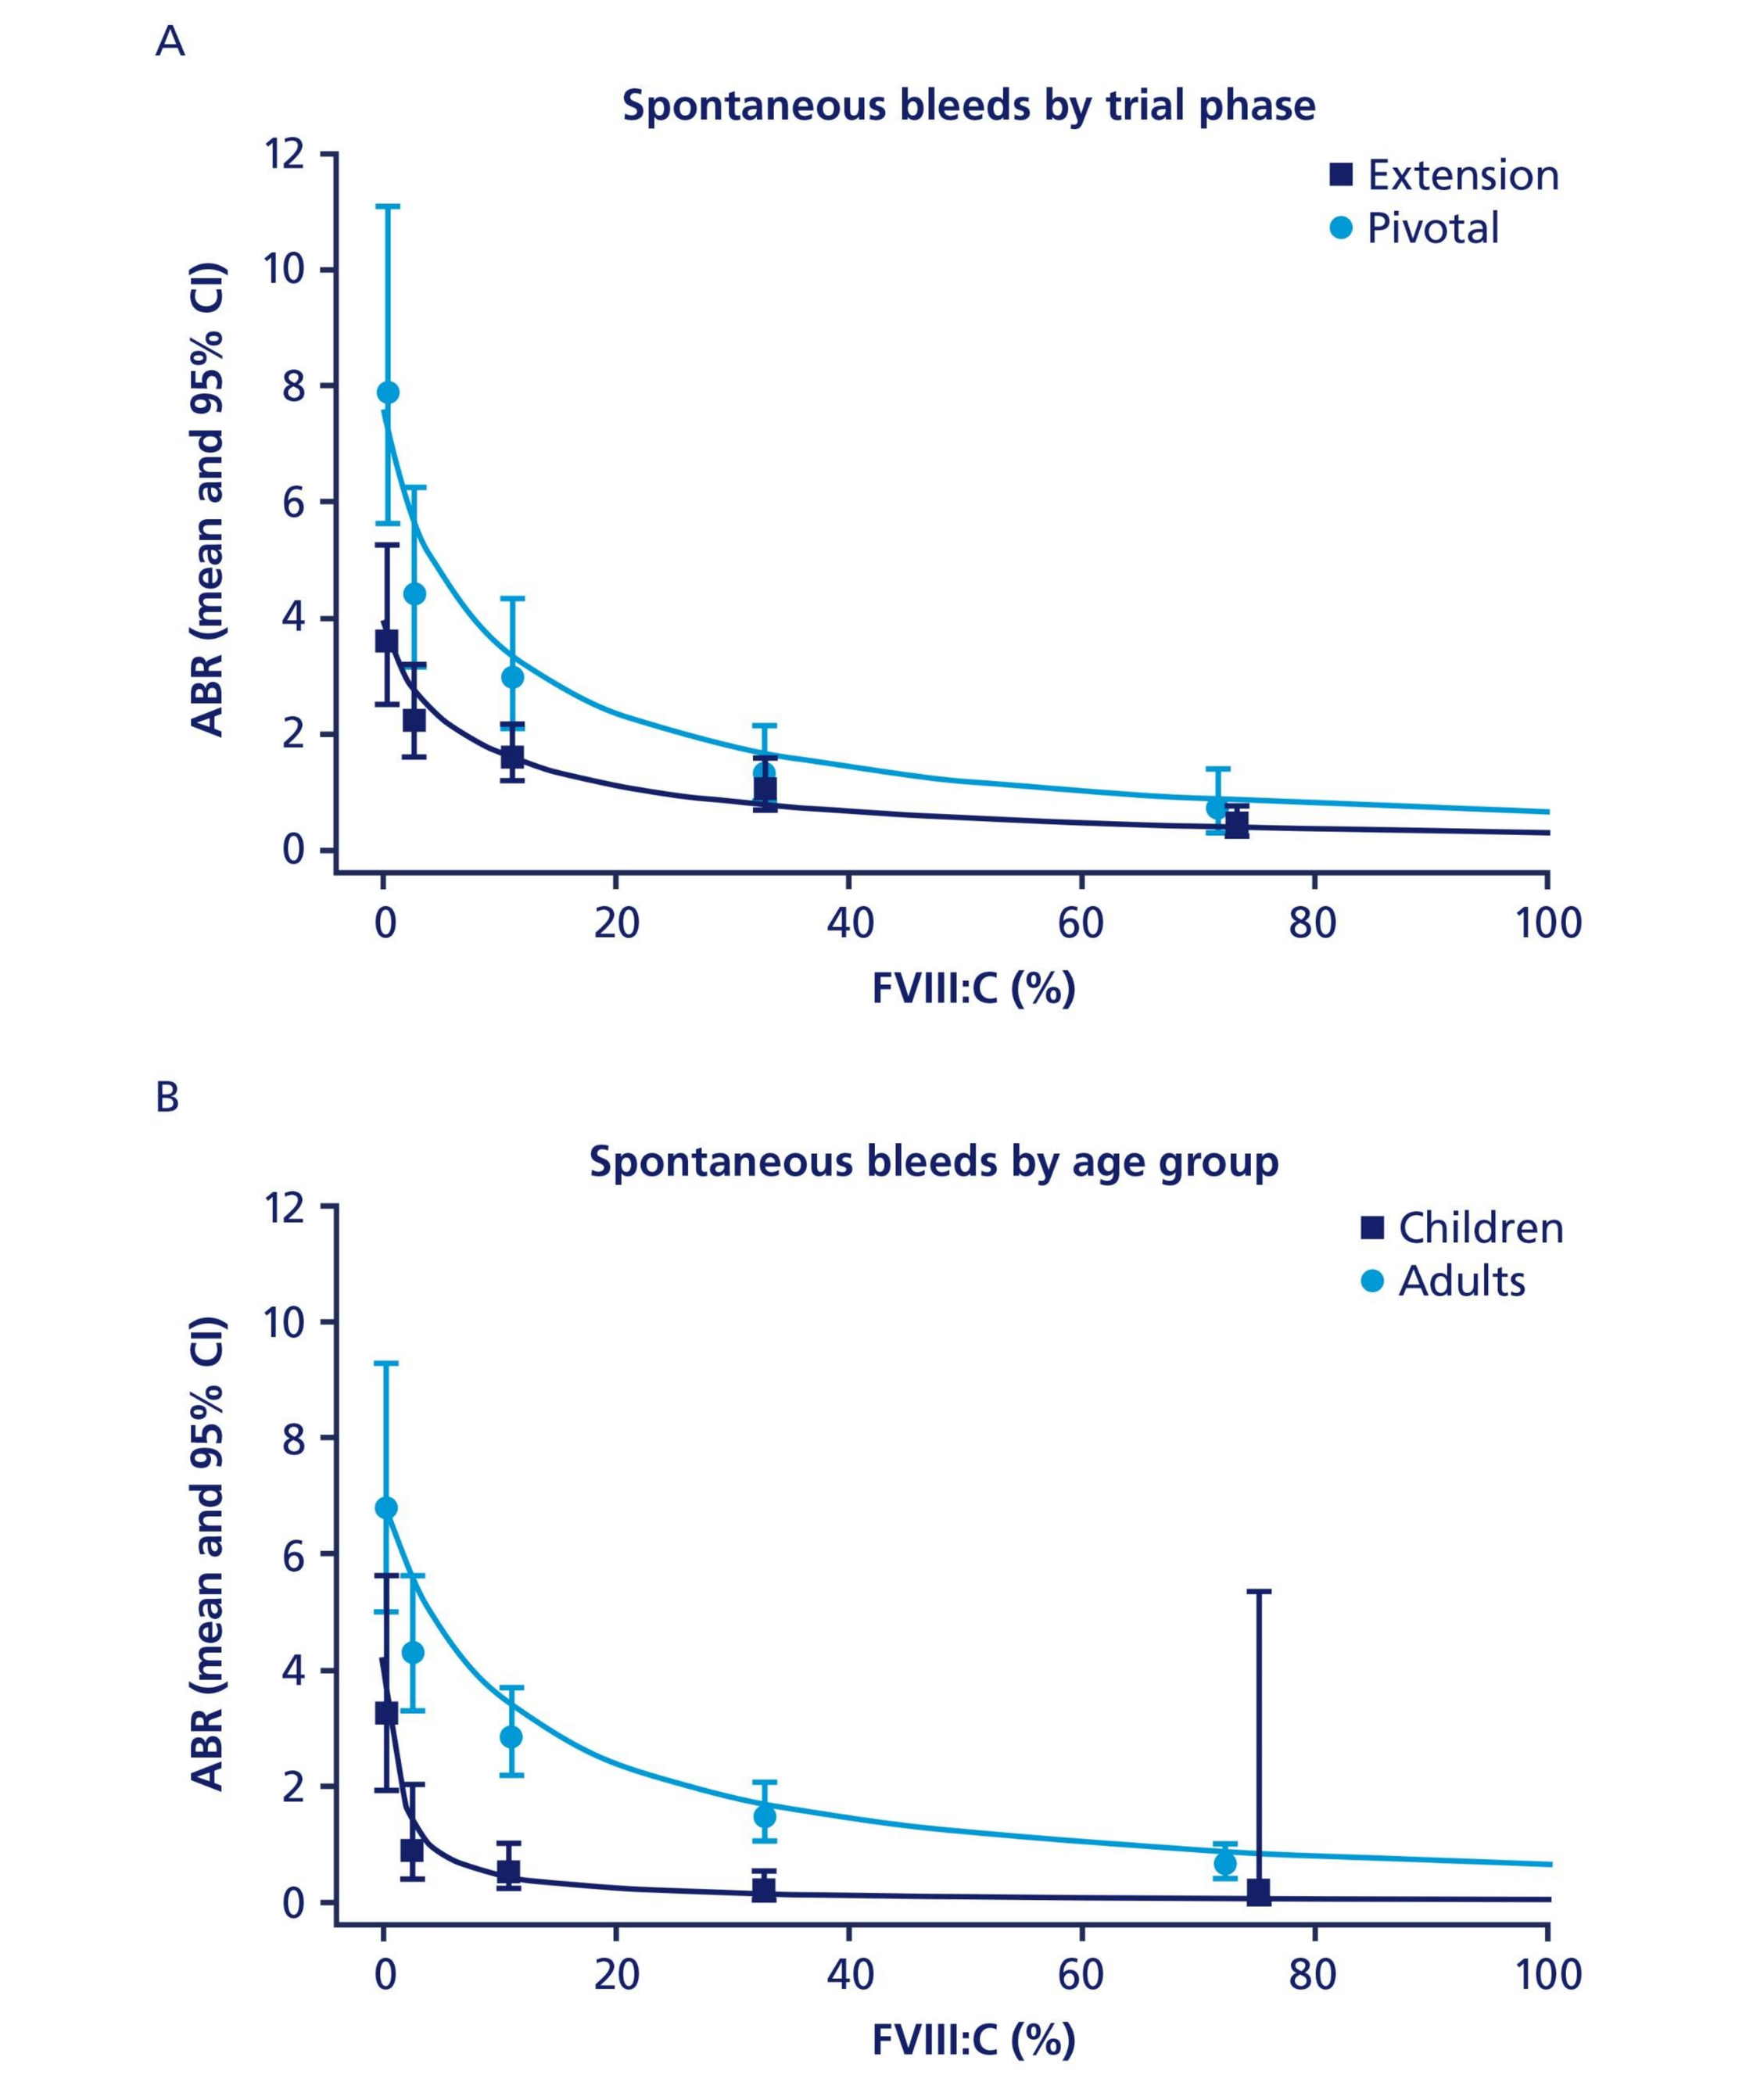
A
Spontaneous bleeds by trial phase
Extension
Pivotal
ABR (mean and 95% CI)
0
2
4
6
8
10
12
0
20
40
60
80
100
FVIII:C (%)
B
Spontaneous bleeds by age group
Children
Adults
ABR (mean and 95% CI)
0
2
4
6
8
10
12
0
20
40
60
80
100
FVIII:C (%)

ABR, annualized bleeding rate; CI, confidence interval; FVIII:C, FVIII coagulation activity.

Table 1 FVIII:C ranges and ABR.

FVIII:C[a]	ABR (95% CI)				
	Analyses population				
	Overall	Trial phase		Age	
		Pivotal	Extension	Adults	Children
0–1%	5.74 (4.42–7.5)	8.13 (5.78–11.5)	3.72 (2.6–5.41)	7.1 (5.24–9.71)	3.27 (1.93–5.63)
>1–5%	3.29 (2.57–4.25)	4.42 (3.15–6.28)	2.26 (1.62–3.2)	4.33 (3.34–5.67)	0.9 (0.42–2.02)
>5–20%	2.23 (1.76–2.86)	3.0 (2.10–4.35)	1.61 (1.19–2.18)	2.86 (2.21–3.72)	0.52 (0.25–1.02)
>20–50%	1.17 (0.85–1.60)	1.33 (0.82–2.14)	1.05 (0.7–1.6)	1.48 (1.06–2.09)	0.21 (0.05–0.54)
>50%	0.48 (0.29–0.74)	0.62 (0.25–1.25)	0.43 (0.23–0.74)	0.6 (0.36–0.93)	0.22 (0.01–5.35)

ABR, annualized bleeding rate; CI, confidence interval; FVIII:C, FVIII coagulation activity.
[a]Each patient's time within the trial is divided into different FVIII:C ranges, based on the diary-recorded dosing pattern and taking into account contributions from the last 3 doses.

Limitations

- Patient dosing patterns, including timing of dosing, were adaptive to the patient's individual needs.
- The judgement of when a treatment-requiring bleed occurs is subjective.

References

1. Lentz S, *et al*. *Haemophilia* 2013;19:691–697.
2. Kulkarni R, *et al*. *Haemophilia* 2013;19:698–705.
3. Ozelo M, *et al*. *Haemophilia* 2015;21:e436–e439.

Conflict of interest disclosure

AVG, **HA**, **IM**, and **RVO** are all full-time employees of Novo Nordisk A/S.
NT is a full-time employee of Novo Nordisk Health Care.

The trials were sponsored by Novo Nordisk and are registered with ClinicalTrials.gov (NCT00840086, NCT01138501, and NCT00984126). The authors acknowledge the medical writing assistance of Mark Simmonds (PAREXEL).
Presented at the World Federation of Hemophilia (WFH) World Congress, July 24–28, 2016, Orlando, FL, USA.

An electronic version of the poster can be viewed by scanning the Quick Response (QR) code. The QR code is intended to provide scientific information for personal use only. The poster should not be altered and may not be reproduced without permission from the authors.

PosterSessionOnline.com
Poster
Presented at:
WFH 2016 WORLD CONGRESS

DOI: 10.3252/pso.eu.WFH2016.2016

Hemophilia - clinical
Harley Souter
91--PP-T
WFH2016