Poster 86

Characteristics of Pediatric Previously Treated Patients With Severe Hemophilia A Aged < 12 Years Experiencing No Bleeds During a 6-Month Prophylactic Treatment Regimen With Pegylated Recombinant Factor VIII

Eric Mullins,[1] Oleksandra Stasyshyn,[2] Diana Osman,[3] Tung T. Wynn,[4] Werner Engl,[5] Borislava Pavlova,[5] Jennifer Doralt,[5] and Brigitt Abbuehl,[5] for the BAX 855 Pediatric Study Group

[1]Cincinnati Children's Hospital Medical Center, Cincinnati, OH, United States; [2]SI Institute of Blood Pathology and Transfusion Medicine of NAMSU, Lviv, Ukraine; [3]Hospital Tengku Ampuan Rahimah, Klang, Selangor, Malaysia; [4]University of Florida College of Medicine, Gainesville, FL, United States; [5*]Shire, Vienna, Austria

INTRODUCTION

- Subjects with severe hemophilia A (factor VIII [FVIII] <1% of normal) experience frequent bleeding which can be prevented by prophylaxis with FVIII replacement therapy.[1]
- ADYNOVATE, a polyethylene glycol pegylated, full-length, recombinant FVIII (rFVIII), was built to extend FVIII half-life on the manufacturing platform of the full-length, unmodified rFVIII rAHF-PFM (ADVATE).
- Due to controlled pegylation, the FVIII half-life of ADYNOVATE was extended by up to 1.5 times compared to ADVATE while maintaining the integrity of the ADVATE protein.[2]
- ADYNOVATE demonstrates preclinical, structural, and functional characteristics comparable to ADVATE.[3,4]
- This Phase 3, prospective, uncontrolled, multicenter, open-label study evaluated the safety, efficacy, and pharmacokinetics (PK) of ADYNOVATE during prophylaxis in previously treated children < 12 years.

OBJECTIVE

- The characteristics of patients treated prophylactically with ADYNOVATE who did not experience bleeding episodes in the pediatric trial were analyzed with respect to:
 - pharmacokinetic profile,
 - pre-study annualized bleeding rates (ABRs)
 - target joint status at screening
 - preexisting arthropathy, and
 - blood group type.
- The results were compared to the characteristics of subjects in the prophylaxis treatment arm who reported at least 1 bleeding episode.

METHODS

Figure 1: Subject Disposition Flowchart

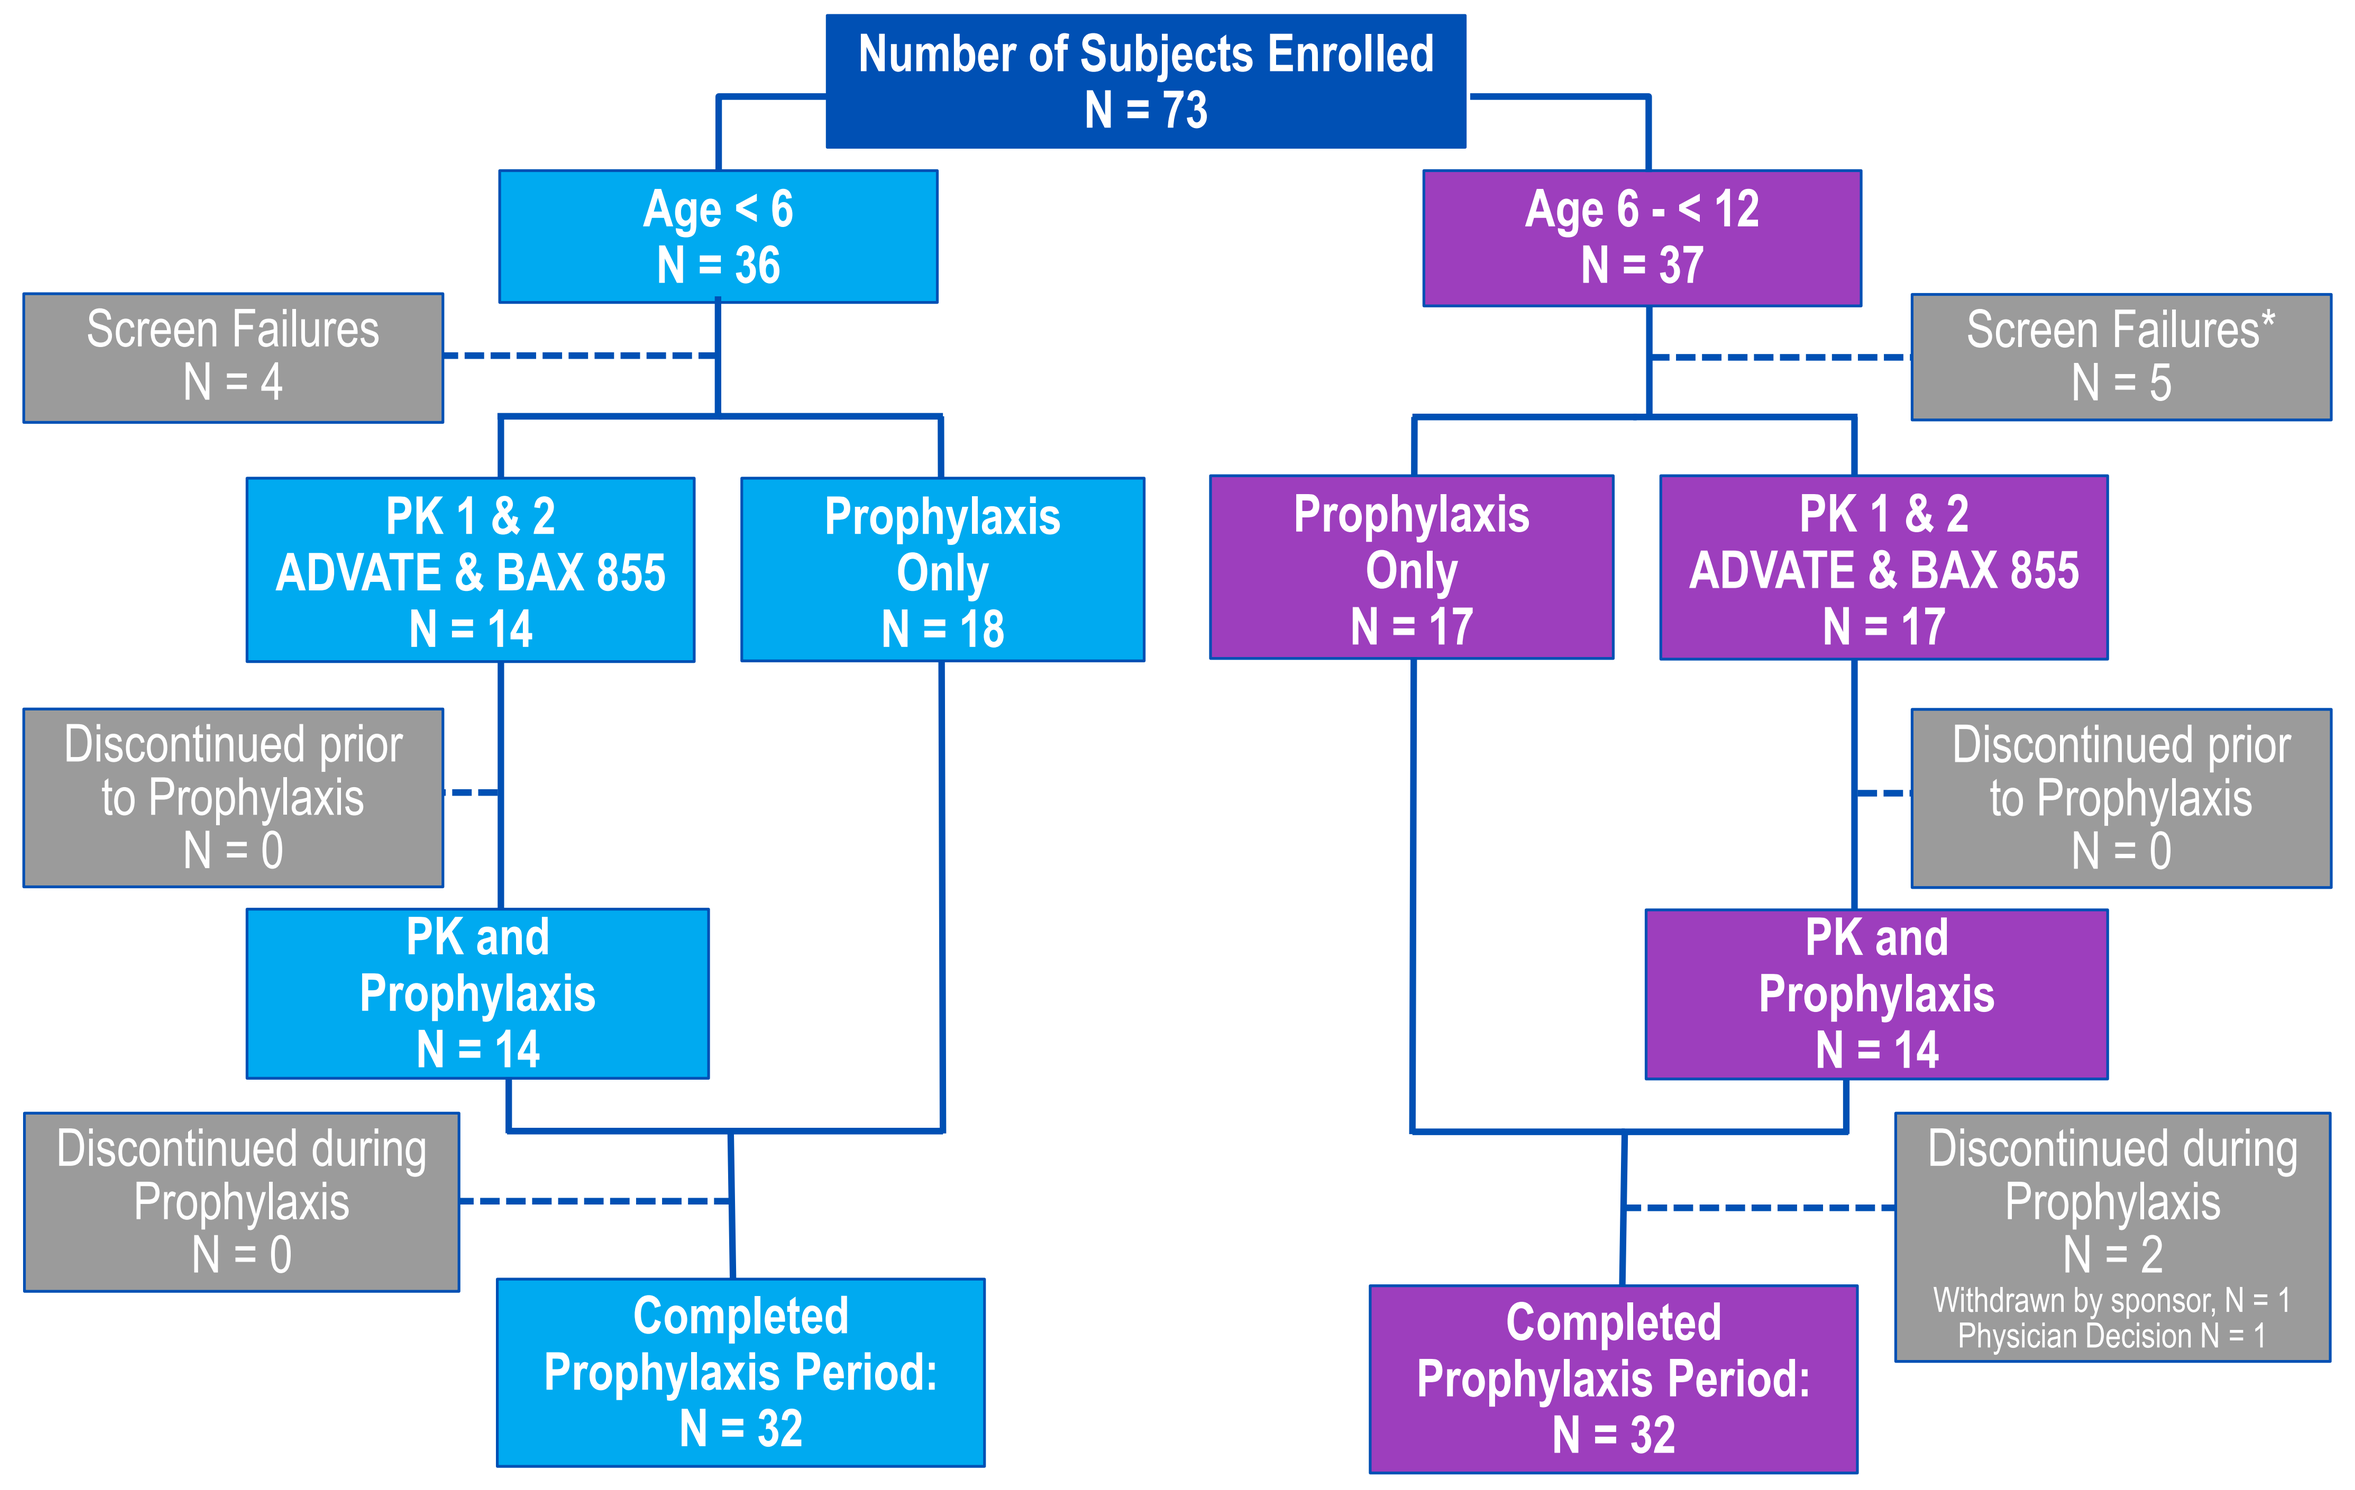
Number of Subjects Enrolled
N = 73
Age < 6
N = 36
Age 6 - < 12
N = 37
Screen Failures
N = 4
Screen Failures*
N = 5
PK 1 & 2
ADVATE & BAX 855
N = 14
Prophylaxis
Only
N = 18
Prophylaxis
Only
N = 17
PK 1 & 2
ADVATE & BAX 855
N = 17
Discontinued prior
to Prophylaxis
N = 0
Discontinued prior
to Prophylaxis
N = 0
PK and
Prophylaxis
N = 14
PK and
Prophylaxis
N = 14
Discontinued during
Prophylaxis
N = 0
Discontinued during
Prophylaxis
N = 2
Withdrawn by sponsor, N = 1
Physician Decision N = 1
Completed
Prophylaxis Period:
N = 32
Completed
Prophylaxis Period:
N = 32

*Two subjects counted as Screen Failures were later enrolled (Unique Subject ID 261202-113001 and 261202-511003).

- This study was conducted in previously treated pediatric patients who had no history of inhibitors
- 73 subjects were enrolled in the study, 66 were dosed.
- There were 2 age cohorts with the following age ranges: < 6 years (32 subjects) and 6 to < 12 years (34 subjects).
- Subjects were enrolled to receive twice weekly prophylactic treatment with 50 ± 10 IU/kg of ADYNOVATE over a period of 6 months or at least 50 exposure days, whichever occurred last.
- Baseline demographics, target joint and arthropathy status at screening, and ABO blood types were analyzed.

WFH World Congress; July 24–28, 2016; Orlando, USA

RESULTS

Baseline Demographics

- No trends were observed related to factors such as race, weight, or BMI.

	No Bleeds N = 24	At Least One Bleed N = 41
Race		
Asian	8 (33.3)	9 (22.0)
Black or African American	2 (8.3)	2 (4.9)
White	14 (58.3)	28 (68.3)
Other	0 (0.0%)	1 (2.4)
Multiple	0 (0.0%)	1 (2.4)
Weight in kg (Mean [SD])	22.67 (7.942)	24.32 (9.069)
BMI in kg/m² (Mean [SD])	16.42 (2.212)	16.42 (2.129)

Subject Characteristics

Gene Mutations

- In the majority of subjects, no known gene mutation result was available or similar.
- Due to the lack of available data, no conclusion can be drawn regarding gene mutations.

	No Bleeds N = 25	At Least One Bleed N = 41
Gene Mutation Available		
Yes	9 (36.0)	18 (43.9)
Frameshift Mutation	1 (4.0)	1 (2.4)
Inversion Intron 1	1 (4.0)	1 (2.4)
Inversion Intron 22	4 (16.0)	6 (14.6)
Missense Mutation	0 (0.0)	1 (2.4)
Point Mutation	2 (8.0)	8 (19.5)
Other	1 (4.0)	1 (2.4)
No	16 (64.0)	22 (53.7)
Missing	0 (0.0)	1 (2.4)

ABO Blood Typing

- A total of 57 study participants underwent ABO blood group determination.
- 40.0% of subjects with no bleeds and 31.7% with at least one bleed had blood group type A; similarly, 36.0% of subjects with no bleeds and 26.8% of subjects with at least one bleed had blood group type O.
- Blood type did not appear to have an effect on bleeding occurrence.

Blood Type	No Bleeds (N = 25)	At Least One Bleed (N = 41)
A	10 (40%)	13 (31.7%)
B	5 (20%)	6 (14.6%)
AB	1 (4%)	2 (4.9%)
0 (null)	9 (36%)	11 (26.8%)
Unknown	0 (0.0%)	8 (19.5%)
Missing	0 (0.0%)	1 (2.4%)

Bleeding Episodes

- Over 6 months of prophylactic treatment, the number of subjects experiencing no bleeding episode was similar between age cohorts: 40.6% (13/32) in subjects < 6 years 35.3% (12/34) in subjects 6 to < 12 years and 37.9% (25/66) overall.
- 72.7% of subjects (48/66) had no bleeds in any joint; in younger children, 78.1% (25/32) and in older childen, 67.6% (23/34).
- 92.4% (61/66 subjects) had no target joint bleeds, 96.9% (31/32) in the younger and 88.2% (30/34) in the older cohort.
- 66.7% (44/66) had no spontaneous or unknown bleeds.

Figure 2: Number of Subjects Without Bleeding Episodes Versus Subjects With At Least 1 Bleeding Episode

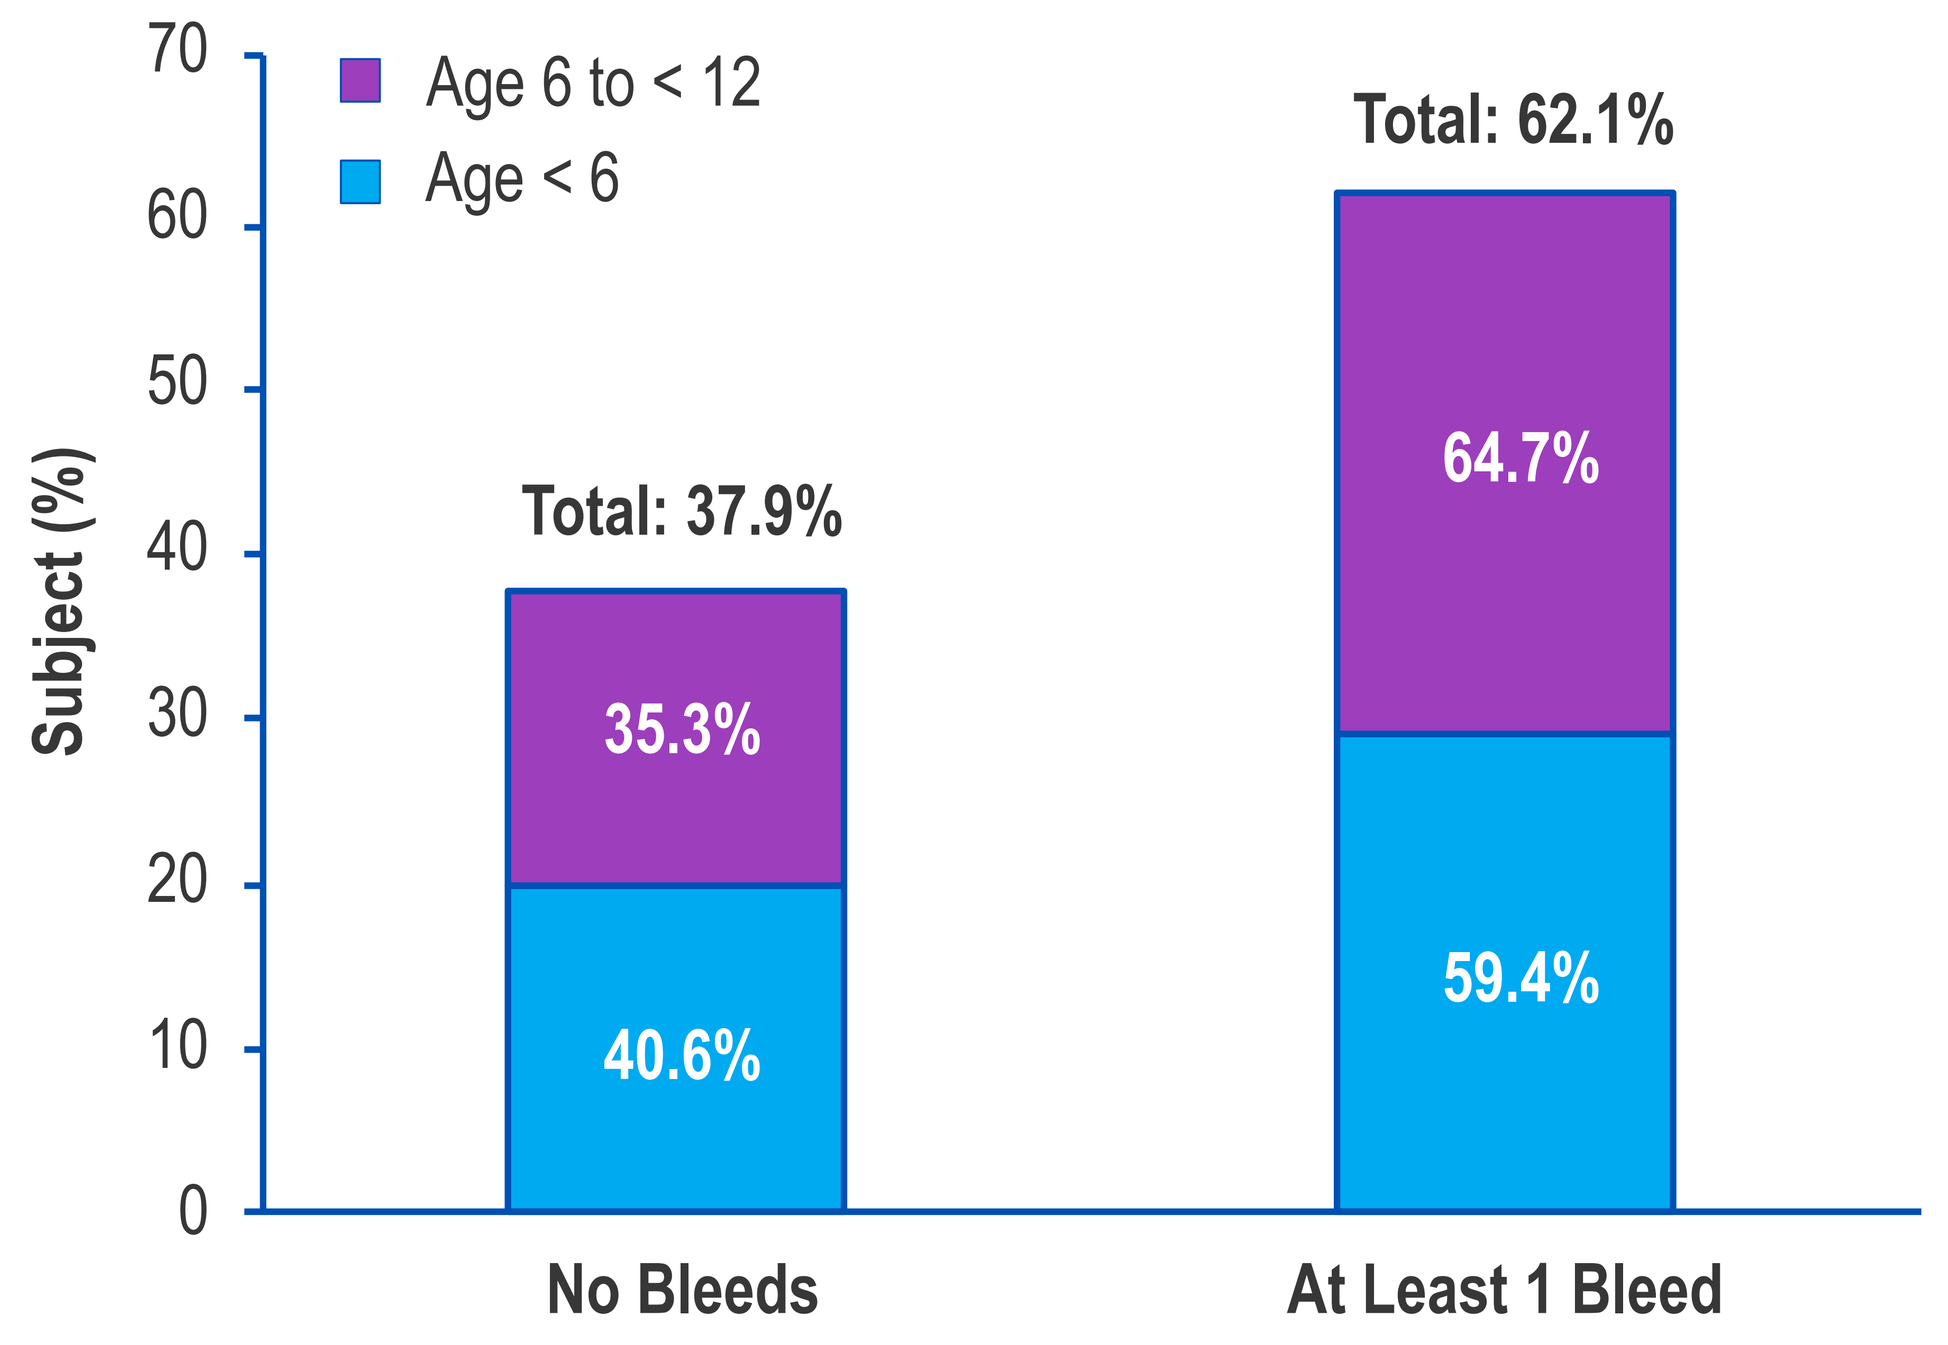
Age 6 to < 12
Age < 6
Subject (%)
70
60
50
40
30
20
10
0
Total: 37.9%
35.3%
40.6%
Total: 62.1%
64.7%
59.4%
No Bleeds
At Least 1 Bleed

Pre-Study Annualized Bleeding Rate (ABR)

- The median pre-study ABR was similar in subjects with and without bleeding episodes (3.0 and 4.0, respectively).
- In the younger cohort, the interquartile range (IQR; Q1, Q3) was 6.00 (0.50, 6.50); among older children, it was 5.00 (1.00, 6.00).
- Of the 5 subjects who received on-demand treatment pre-study, 4 did and only 1 did not experience a bleeding episode during the study.

Target Joints

- A target joint was defined as single joint (ankle, knee, hip, or elbow) with ≥ 3 spontaneous bleeding episodes in any consecutive 6-month period.
- The majority of subjects 78.8% (52/66) had no target joints at screening.
- 24.4% of subjects with bleeding and 16.0% of patients without bleeding had 1 or more target joints at screening.
- No target joints were reported in 75.6% of subjects with bleeding episodes, compared to 84.0% of those without bleeding episodes.
- In the younger cohort, 9.4% (3/32) subjects had one target joint compared with 32.4% (11/34) in the older cohort who had at least 1 target joint at screening.
- Patients with at least 1 target joint were more likely to have at least one bleed compared to those with no target joint at screening.

DISCLOSURES

EM, OS, DO, and TTW were investigators in this study, Baxalta US, Inc. and Baxalta Innovations GmbH; Baxalta, now part of Shire. *WE, BP, JD and BA are full-time employees of Baxalta, (Baxalta Innovations GmbH), now part of Shire.

The study was registered at www.clinicaltrials.gov as NCT02210091 and at www.clinicaltrialsregister.eu under EudraCT Number 2014 000742 30. The study was funded by Baxalta US, Inc, now part of Shire.

Figure 3: Target Joints in Patients Without Bleeding Compared to Patients With at Least 1 Bleeding Episode

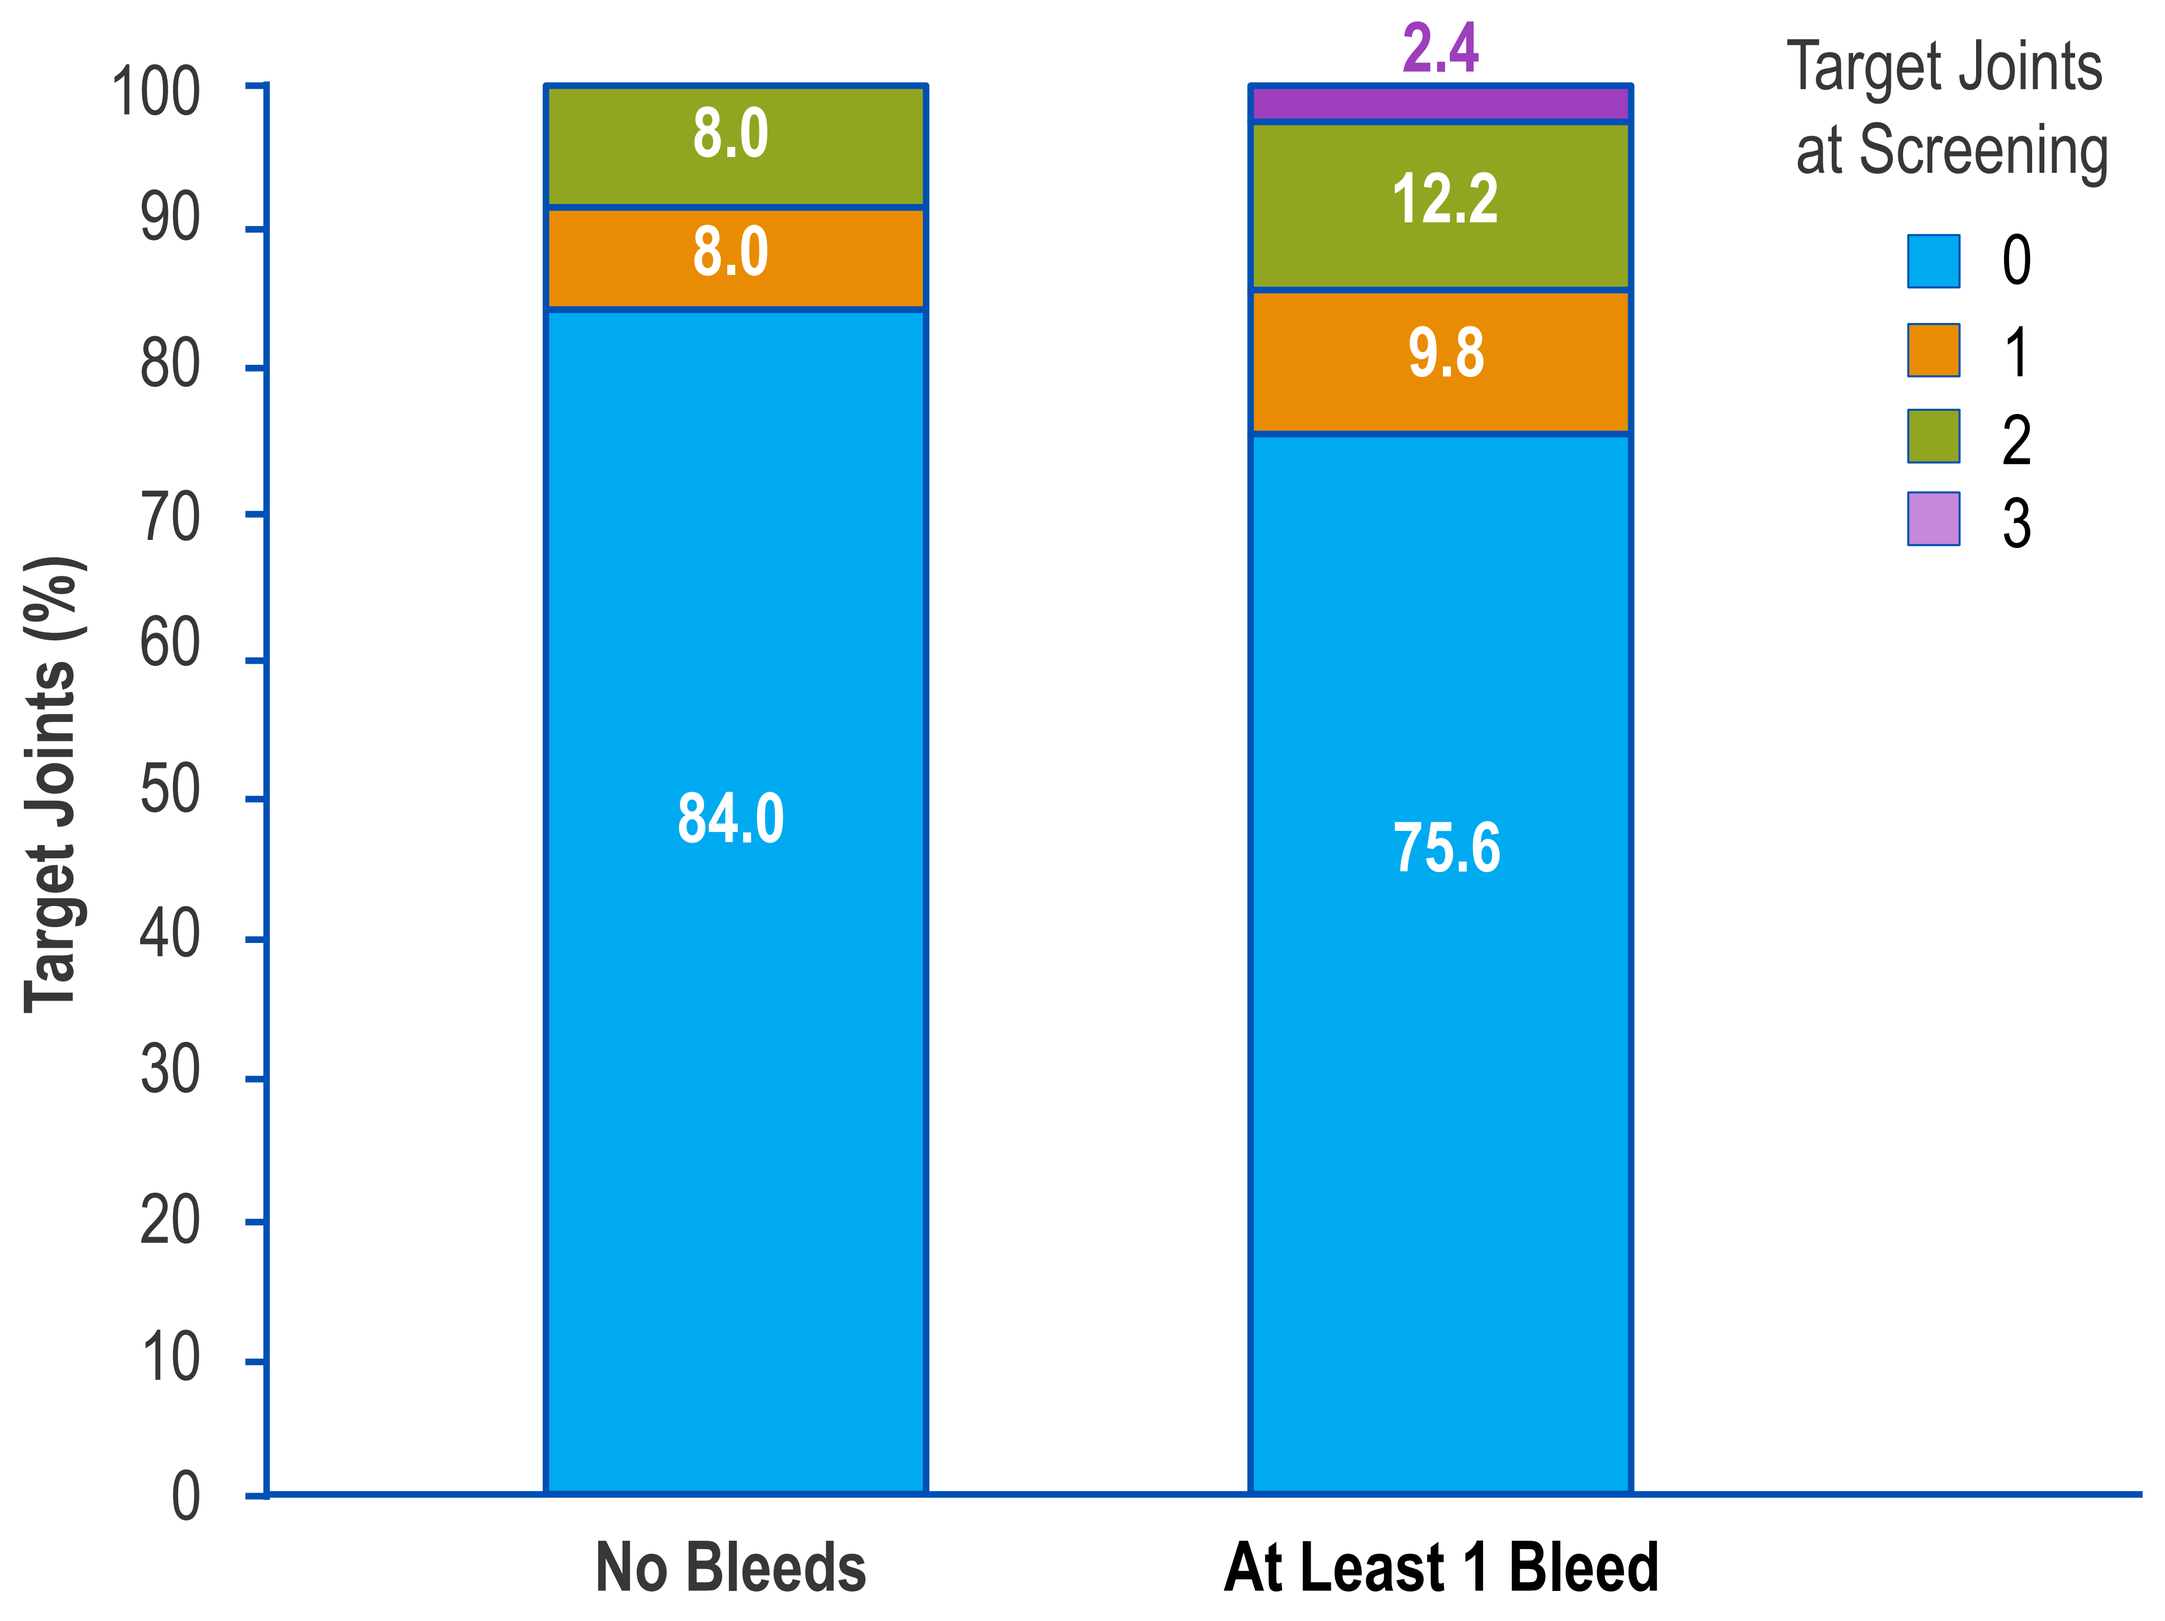
Target Joints (%)
100
90
80
70
60
50
40
30
20
10
0
8.0
8.0
84.0
2.4
12.2
9.8
75.6
No Bleeds
At Least 1 Bleed
Target Joints at Screening
0
1
2
3

Arthropathy

- 3/34 (8.8%) subjects in the older cohort and no subject in the younger cohort had arthropathy at screening.
- 2/25 (8.0%) subjects with no bleeds and 1/41 (2.4%) with bleeds had hemophilic arthropathy.
- No conclusion can be drawn regarding the impact on bleeding given the low sample size of n = 3 with hemophilic arthropathy at screening.

SUMMARY

- Pre-study ABR was similar in subjects who did not experience bleeding compared to patients with bleeding.
- No trends were observed related to the type of gene mutation or blood type.
- The percentage of subjects having 1 or more target joints at screening was higher in subjects experiencing at least one bleed than in subjects with no bleeds.
- The number of subjects experiencing no bleeding episodes was similar between age cohorts: subjects < 6 years old versus subjects 6 to < 12 years old.

CONCLUSIONS

- While most characteristics of subjects on prophylaxis with and without bleeding episodes were similar, subjects with bleeding episodes had more target joints at screening.
- The majority of subjects did not experience joint, target joint, or spontaneous bleeds during treatment with ADYNOVATE.

REFERENCES

1. Srivastava A, et al. Guidelines for the management of hemophilia. *Haemophilia*. 2013;19:e1–e47.
2. Bevan D, et al. A Phase 1 study of safety and pharmacokinetics (PK) of BAX 855, a longer acting pegylated full-length recombinant Factor VIII (PEG-rFVIII), in patients (pts) with severe haemophilia A. *Haemophilia.* 2013;(s2):Po053, pp.32.
3. Turecek PL, et al. BAX 855, a PEGylated rFVIII with prolonged half-life. *Hämostaseologie*. 2012;32(1):S29-S38.
4. Konkle BA, et al. Pegylated, full-length, recombinant factor VIII for prophylactic and on-demand treatment of severe hemophilia A. *Blood* 2015;126(9): 1078-1085..

PosterSessionOnline.com
Poster Presented at:
WFH 2016 World Congress
DOI: 10.3252/pso.eu.WFH2016.2016
Hemophilia - clinical
Jennifer Doralt
86--PP-T
WFH2016