Comparison of the pharmacokinetic parameters of a plasma-derived VWF/FVIII concentrate (Voncento®) in adult/adolescent and pediatric subjects with von Willebrand disease (SWIFT-VWD and SWIFTLY-VWD study)

T Lissitchkov[1], E Buevich[2], K Kuliczkowski[3], O Stasyshyn[4], MH Cerqueira[5], A Klukowska[6], G Auerswald[7], C Djambas Khayat[8], G Iosova[9], I Ramasheuskaya[10], MJ Lopez Ruano[11], C Joch[12], W Seifert[12], T Rogosch[12]

[1]Specialized Hospital for Active Treatment (SHAT) “Joan Pavel”, Sofia, Bulgaria, [2]Altay State Medical University of Roszdrav, Barnaul, Russian Federation, [3]Independent Public Clinical Hospital No 1, Wroclaw, Poland, [4]Institute of Blood Pathology and Transfusion Medicine, Academy of Medical Sciences of Ukraine, Lviv, Ukraine, [5]Institute of Hematology, Rio de Janeiro, Brazil, [6]Department of Pediatrics, Hematology and Oncology, Warsaw Medical University, Warsaw, [7]Prof. Hess Kinderklinik, Klinikum Bremen-Mitte, Bremen, Germany, [8]Hemophilia Care Center, Hotel Dieu de France Hospital, Beirut, Lebanon, [9]Institute for Hematology and Infusion, Tbilisi, Georgia, [10]Republican Scientific and Practical Centre of Children Oncology and Hematology, Gomel, Belarus, [11]Hematology, Hospital Roosevelt, Guatemala, Guatemala, [12]Clinical Development, CSL Behring, Marburg, Germany

Introduction

Voncento® is a plasma-derived, high-concentration, low-volume, high-purity concentrate, which contains a high level of high-molecular-weight multimers and a VWF:FVIII ratio of ~2.4:1.
The SWIFT (“**S**tudies with von **Wi**llebrand factor/**F**ac**t**or VIII”) program is evaluating Voncento® in hemophilia A and VWD patients in accordance with the European clinical and pediatric guidelines. One aim of the two open-label studies SWIFT-VWD and SWIFTLY-VWD was to compare the pharmacokinetic (PK) parameters in pediatric subjects aged 0 to <12 years with adult/ adolescent subjects.

Study design

Subjects with severe VWD received Voncento® as a single bolus infusion of 80 IU VWF:RCo/kg body weight on Day 1 and 180 (n=14: pediatrics, n=12: adults/adolescents). PK parameters for VWF and FVIII were derived from plasma concentration values collected prior to dosing and at 0.5, 4, 8, 24, and 48 h after infusion. PK parameters comprised Incremental recovery (IR), Half-life ($t_{1/2}$), AUC, C_{max}, t_{max}, Mean residence time (MRT), Clearance (CL), and Volume of distribution at steady state (V_{ss}).

PK parameters

adults	VWF:RCo		VWF:Ag		VWF:CB		FVIII:C	
	N	median (range)	N	median (range)	N	median (range)	N	median (range)
IR [(IU/mL)/(IU/kg)]	12	0.017 (0.012-0.021)	12	0.018 (0.013-0.022)	12	0.022 (0.015-0.025)	12	0.027 (0.016-0.036)
$t_{1/2}$ [h]	8	11.53 (6.05-35.10)	12	18.39 (11.41-27.01)	12	14.54 (9.36-25.10)	10	23.65 (7.69-57.48)
MRT [h]	8	13.25 (8.59-25.45)	12	24.57 (15.28-33.60)	12	18.74 (11.61-28.57)	10	36.57 (15.62-85.14)
AUC_{0-72} [h*IU/mL]	12	14.46 (8.56-37.99)	12	33.1 (22.65-64.68)	12	24.32 (14.83-41.14)	11	27.85 (13.15-66.82)
C_{max} [IU/mL]	12	1.48 (0.93-3.36)	12	2.04 (1.52-3.66)	12	1.6 (1.04-2.66)	12	1 (0.57-1.32)
CL [mL/(h*kg)]	12	6.16 (3.06-9.32)	12	3.74 (2.61-4.78)	12	3.2 (2.32-4.77)	11	1.28 (0.62-2.47)
V_{SS} [mL/kg]	8	68.3 (44.7-158.0)	12	74 (64.5-128.4)	12	71 (47.5-93.7)	10	47.5 (24.8-72.9)

pediatrics	VWF:RCo		VWF:Ag		VWF:CB		FVIII:C	
	N	median (range)	N	median (range)	N	median (range)	N	median (range)
IR [(IU/mL)/(IU/kg)]	14	0.015 (0.009-0.017)	14	0.014 (0.007-0.022)	14	0.014 (0.009-0.017)	13	0.020 (0.008-0.048)
$t_{1/2}$ [h]	8	11.40 (4.13-22.44)	13	11.12 (7.72-22.36)	13	10.16 (6.08-15.44)	7	19.01 (7.05-32.61)
MRT [h]	8	13.01 (4.36-32.74)	13	13.26 (9.03-31.68)	13	12.50 (7.17-20.96)	7	25.57 (6.63-44.40)
AUC_{0-48} [h*IU/mL]	14	9.50 (3.11-17.71)	14	19.60 (11.71-34.55)	14	15.65 (11.10-25.30)	13	16.10 (1.47-34.82)
C_{max} [IU/mL]	14	1.15 (0.69-1.35)	14	1.67 (1.22-2.50)	14	1.43 (1.13-1.93)	13	0.69 (0.33-1.46)
CL [mL/(h*kg)]	8	7.26 (2.82-17.32)	13	4.98 (2.24-13.13)	13	6.60 (3.66-11.74)	7	2.96 (0.96-26.07)
V_{SS} [mL/kg]	8	93.1 (52.3-135.3)	13	71.1 (54.6-133.5)	13	83.2 (54.7-113.8)	7	76.6 (33.1-172.9)

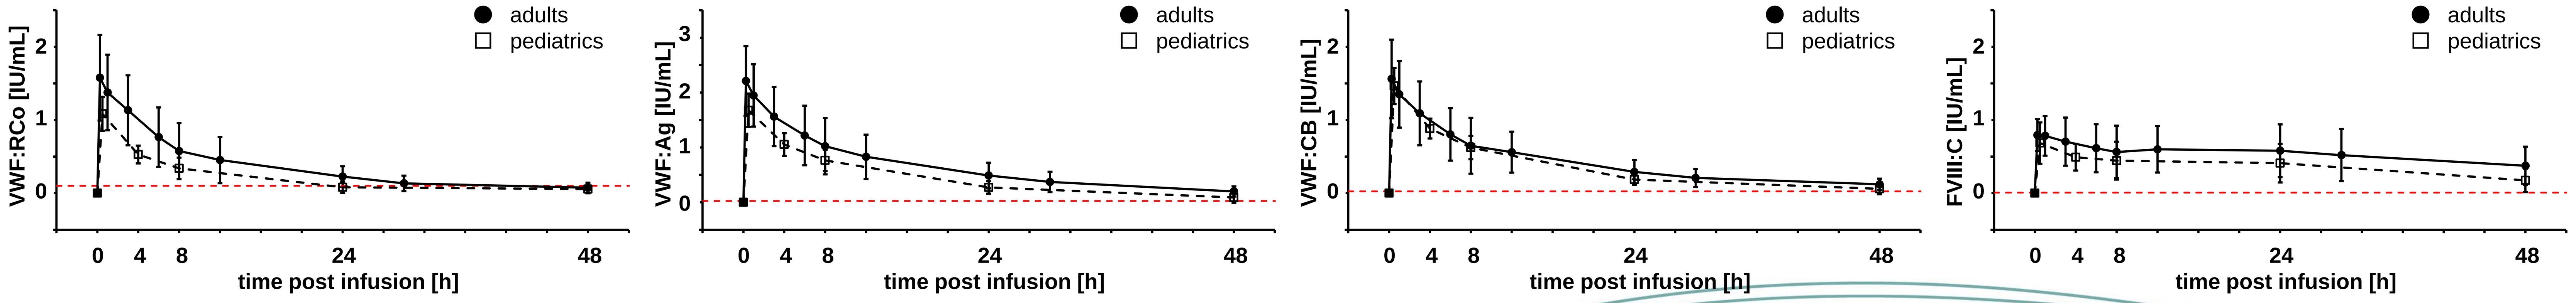
adults
pediatrics
VWF:RCo [IU/mL]
VWF:Ag [IU/mL]
VWF:CB [IU/mL]
FVIII:C [IU/mL]
0 4 8 24 48
time post infusion [h]

Mean (SD) concentration profiles [IU/mL] of baseline-adjusted VWF:RCo, VWF:Ag, VWF:CB, and FVIII:C (adults [N=12], pediatrics [N=14]); red dashed line: lower limit of quantitation (VWF:RCo: 0.1 IU/mL; VWF:Ag, VWF:CB: 0.025 IU/mL; FVIII:C: 0.008 IU/mL)

Results

Overall, pediatric subjects showed a comparable but slightly lower VWF:RCo exposure than adults/adolescents as indicated by a lower IR (median: 0.015 versus 0.017 (IU/mL)/(IU/kg)), slightly shorter $t_{1/2}$ (11.4 versus 11.5 h), faster CL (7.26 versus 6.16 mL/(h*kg)) and higher V_{ss} (93.1 versus 68.3 mL/kg). VWF:Ag, VWF:CB, and FVIII:C showed similar trends with FVIII:C showing longer $t_{1/2}$ (19.0 versus 23.7 h) and a slower clearance (2.96 versus 1.28 mL/(h*kg)) compared to the VWF markers due to a plateau effect that may represent the net effect of decreasing levels of exogenous FVIII, combined with increasing endogenous FVIII levels. PK parameters from the repeat PK were similar to those from initial PK.

Conclusion

Although in general comparable to adults/adolescents, pediatric patients under 12 years of age had higher volumes of distribution and faster clearance for VWF markers as known for almost all factor concentrates used for replacement therapy. Thus, for prophylaxis treatment a higher dose range of 40-80 IU VWF:RCo/kg body weight (adults/adolescents: 25-40 IU VWF:RCo/kg body weight) 1-3 times a week should be considered to compensate for faster elimination and greater volume of distribution in pediatric subjects.

Biotherapies for Life™ CSL Behring

PosterSessionOnline.com
Poster Presented at: WFH 2016 World Congress
DOI: 10.3252/pso.eu.WFH2016.2016
Von Willebrand disease
Tobias Rogosch
185--PP-T
WFH2016