P-T-174

Impact of Mild to Severe Hemophilia on Engagement in Recreational Activities by US Adult Men and Women With Hemophilia B: The Bridging Hemophilia B Experiences, Results and Opportunities Into Solutions (B-HERO-S) Study

Kimberly Baumann,[1] Grace Hernandez,[2] Michelle Witkop,[3] Skye Peltier,[1] Spencer Dunn,[2] Susan Cutter,[4] Neil Frick,[5] Kimberly Haugstad,[6] Neeraj Iyer,[7] David L. Cooper[7]

[1]University of Minnesota Health, Center for Bleeding and Clotting Disorders, Minneapolis, MN; [2]Center for Comprehensive Care and Diagnosis of Inherited Bleeding Disorders, Orange, CA; [3]Munson Medical Center, Traverse City, MI; [4]Hospital of the University of Pennsylvania, Philadelphia, PA; [5]National Hemophilia Foundation, New York, NY; [6]Hemophilia Federation of America, Washington, DC; [7]Novo Nordisk Inc., Plainsboro, NJ.

Objective

- B-HERO-S was designed to better understand the psychosocial impact of hemophilia on men, women, and children with mild-moderate-severe hemophilia B, and in particular, to explore the impact of hemophilia on engagement in activities.

Introduction

- Initiated in 2009, the global HERO program and subsequent 10-country HERO quantitative study sought to investigate the psychosocial issues affecting people with hemophilia (PWH) and those who care for and support them.[1,2]
- Results from HERO highlighted difficulty in managing the pain/disability and sequelae that arise as a consequence of frequent joint bleeding and chronic joint damage.
- As HERO covered a broad patient group, less is known about the psychosocial impact of hemophilia on people with hemophilia B, specifically.
- B-HERO-S was designed in collaboration with health care professionals, advocacy organizations, and patients/caregivers to assess the needs in this population and to address gaps identified by the HERO quantitative study data:
 - Impact on people with mild-moderate hemophilia, including affected women/girls
 - Assessment of the vigorousness and duration of activity participation
 - Treatment modifications to support ongoing participation in activities

Methods

- US adults (either gender, age ≥18 years) with hemophilia B were recruited through 3 patient organizations to complete an approximately 1-hour Internet-based IRB-approved survey available from September 26 through November 3, 2015.
- Surveys assessed the impact of hemophilia on activities, including current, past, and aspirational activities (selected from a list of 35 different activities), duration and vigorousness of current activities, reasons for not currently participating in activities, and specific changes to treatment regimens around activities.

Results

Demographics

- A total of 299 adults participated in the survey; approximately one-third were females.
- Respondent demographics are summarized according to hemophilia severity in **Table 1.**

Table 1 Demographics

Parameter	Total (N=299)	Mild (n=74)	Moderate (n=189)	Severe (n=32)	Inhibitors (n=4)
Gender, n					
Male	213	49	133	27	4
Female	86	25	56	5	0
Age, years					
Median	29	28	29	30	22.5
(Min, max)	(18, 70)	(18, 70)	(18, 70)	(18, 61)	(20, 40)
Treatment, %					
On demand	10	20	7	3	25
Routine infusions	80	63	86	81	75
On demand + extra infusions	9	12	7	13	0

Impact of hemophilia on current activities

- Overall, 98% of respondents reported a negative impact of hemophilia on engagement in recreational activities (**Figure 1**).
 - A "moderate" or "large" impact was reported by 64% of those with mild hemophilia, 93% of those with moderate hemophilia, and 78% of those with severe hemophilia.
 - Greater numbers of participants receiving routine infusions (91%), compared with those receiving on demand (55%) or on demand + extra infusions (58%), reported a "moderate" or "large" impact.
 - Responses were similar between males and females.

Figure 1 Impact on current participation in recreational activities

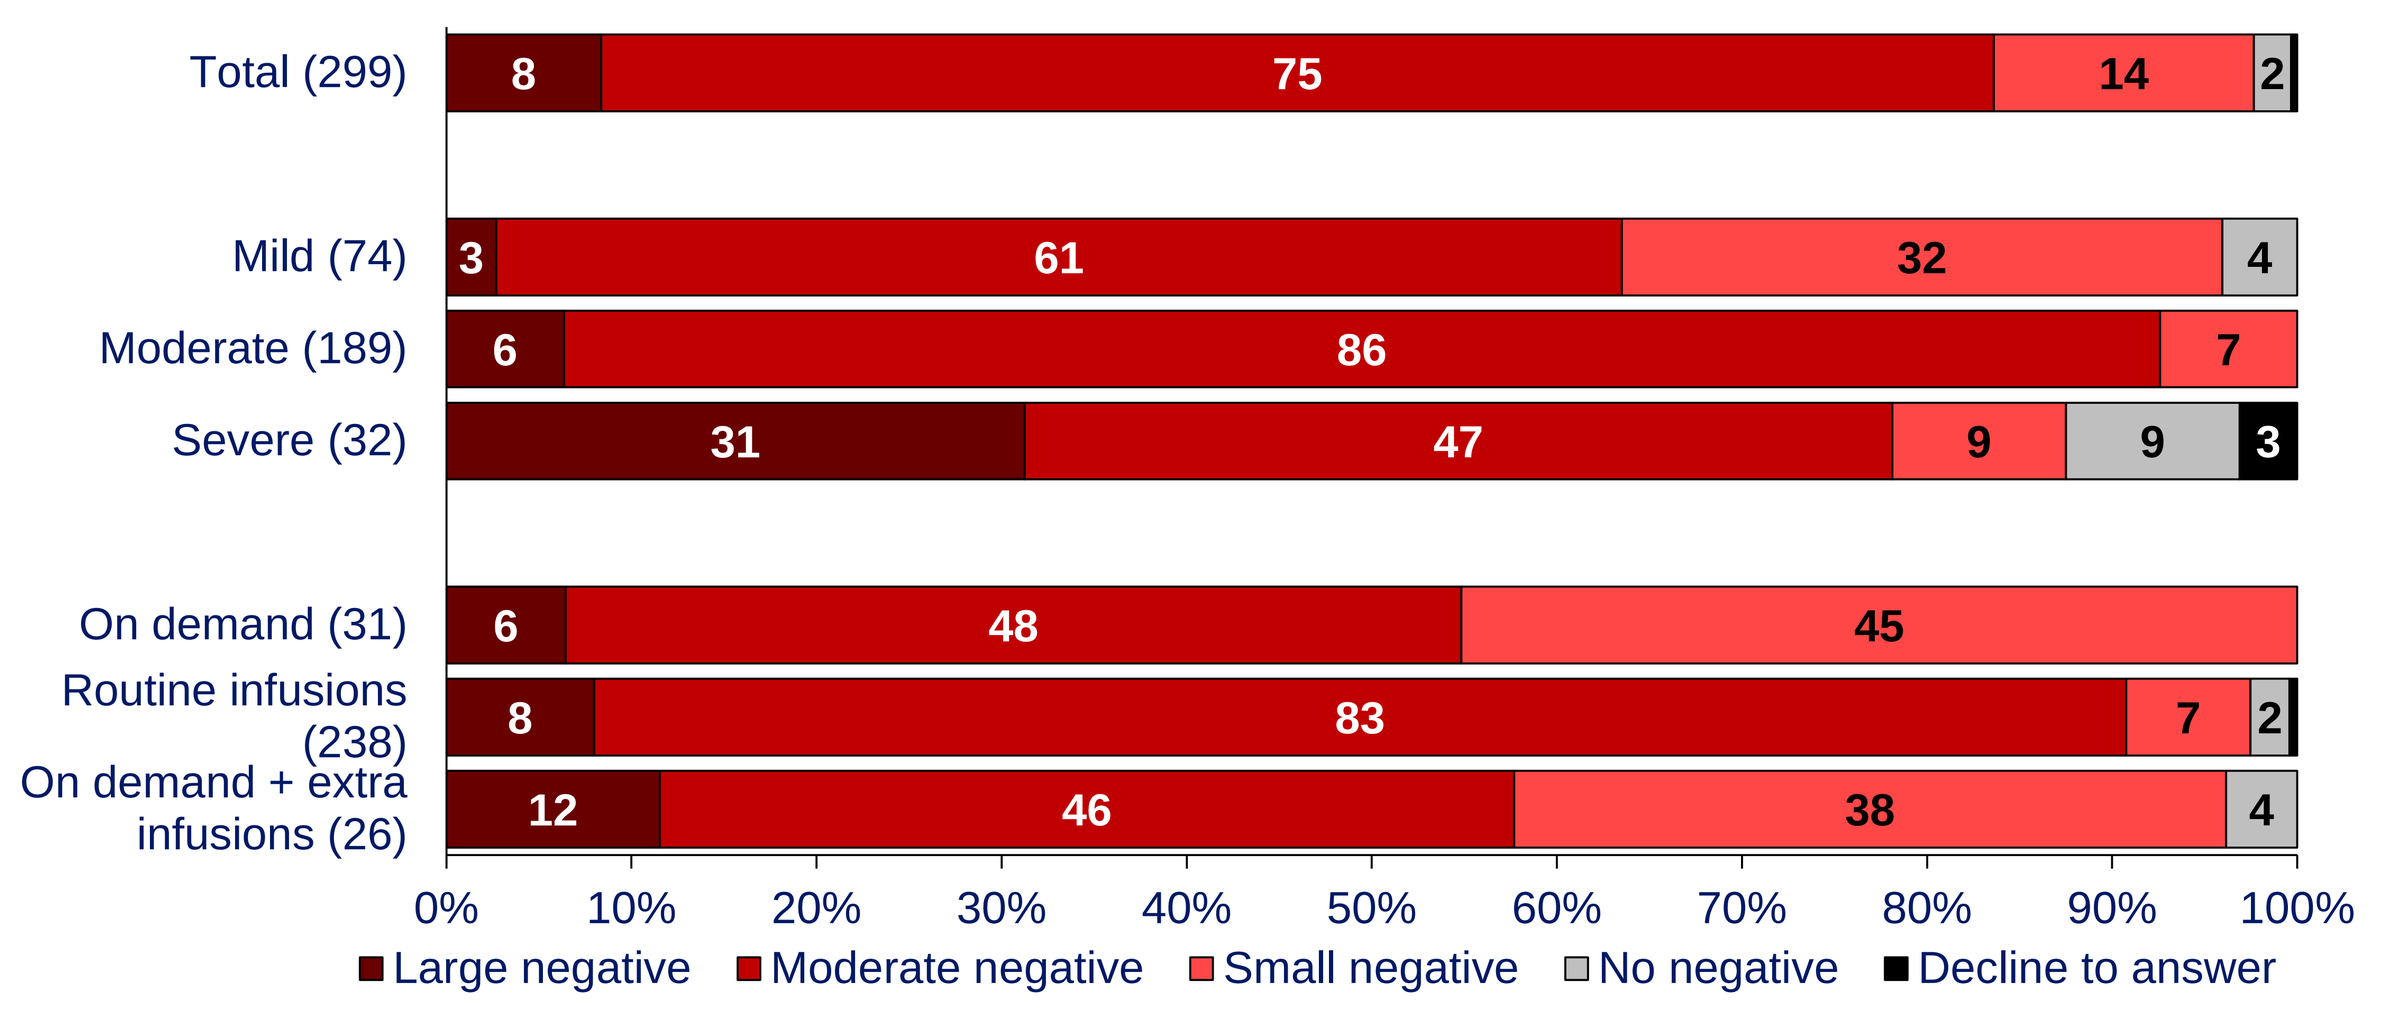

Total (299)
Mild (74)
Moderate (189)
Severe (32)
On demand (31)
Routine infusions (238)
On demand + extra infusions (26)
Large negative
Moderate negative
Small negative
No negative
Decline to answer

Parentheses indicate the numbers of PWH in each category.

- The most common current activities reported were walking (44%), dancing (26%), fishing (19%), and bicycling (16%).
- The intensity (low/moderate/vigorous) and weekly duration of activities are shown in **Figure 2**.

Figure 2 Intensity and duration of current participation in activities

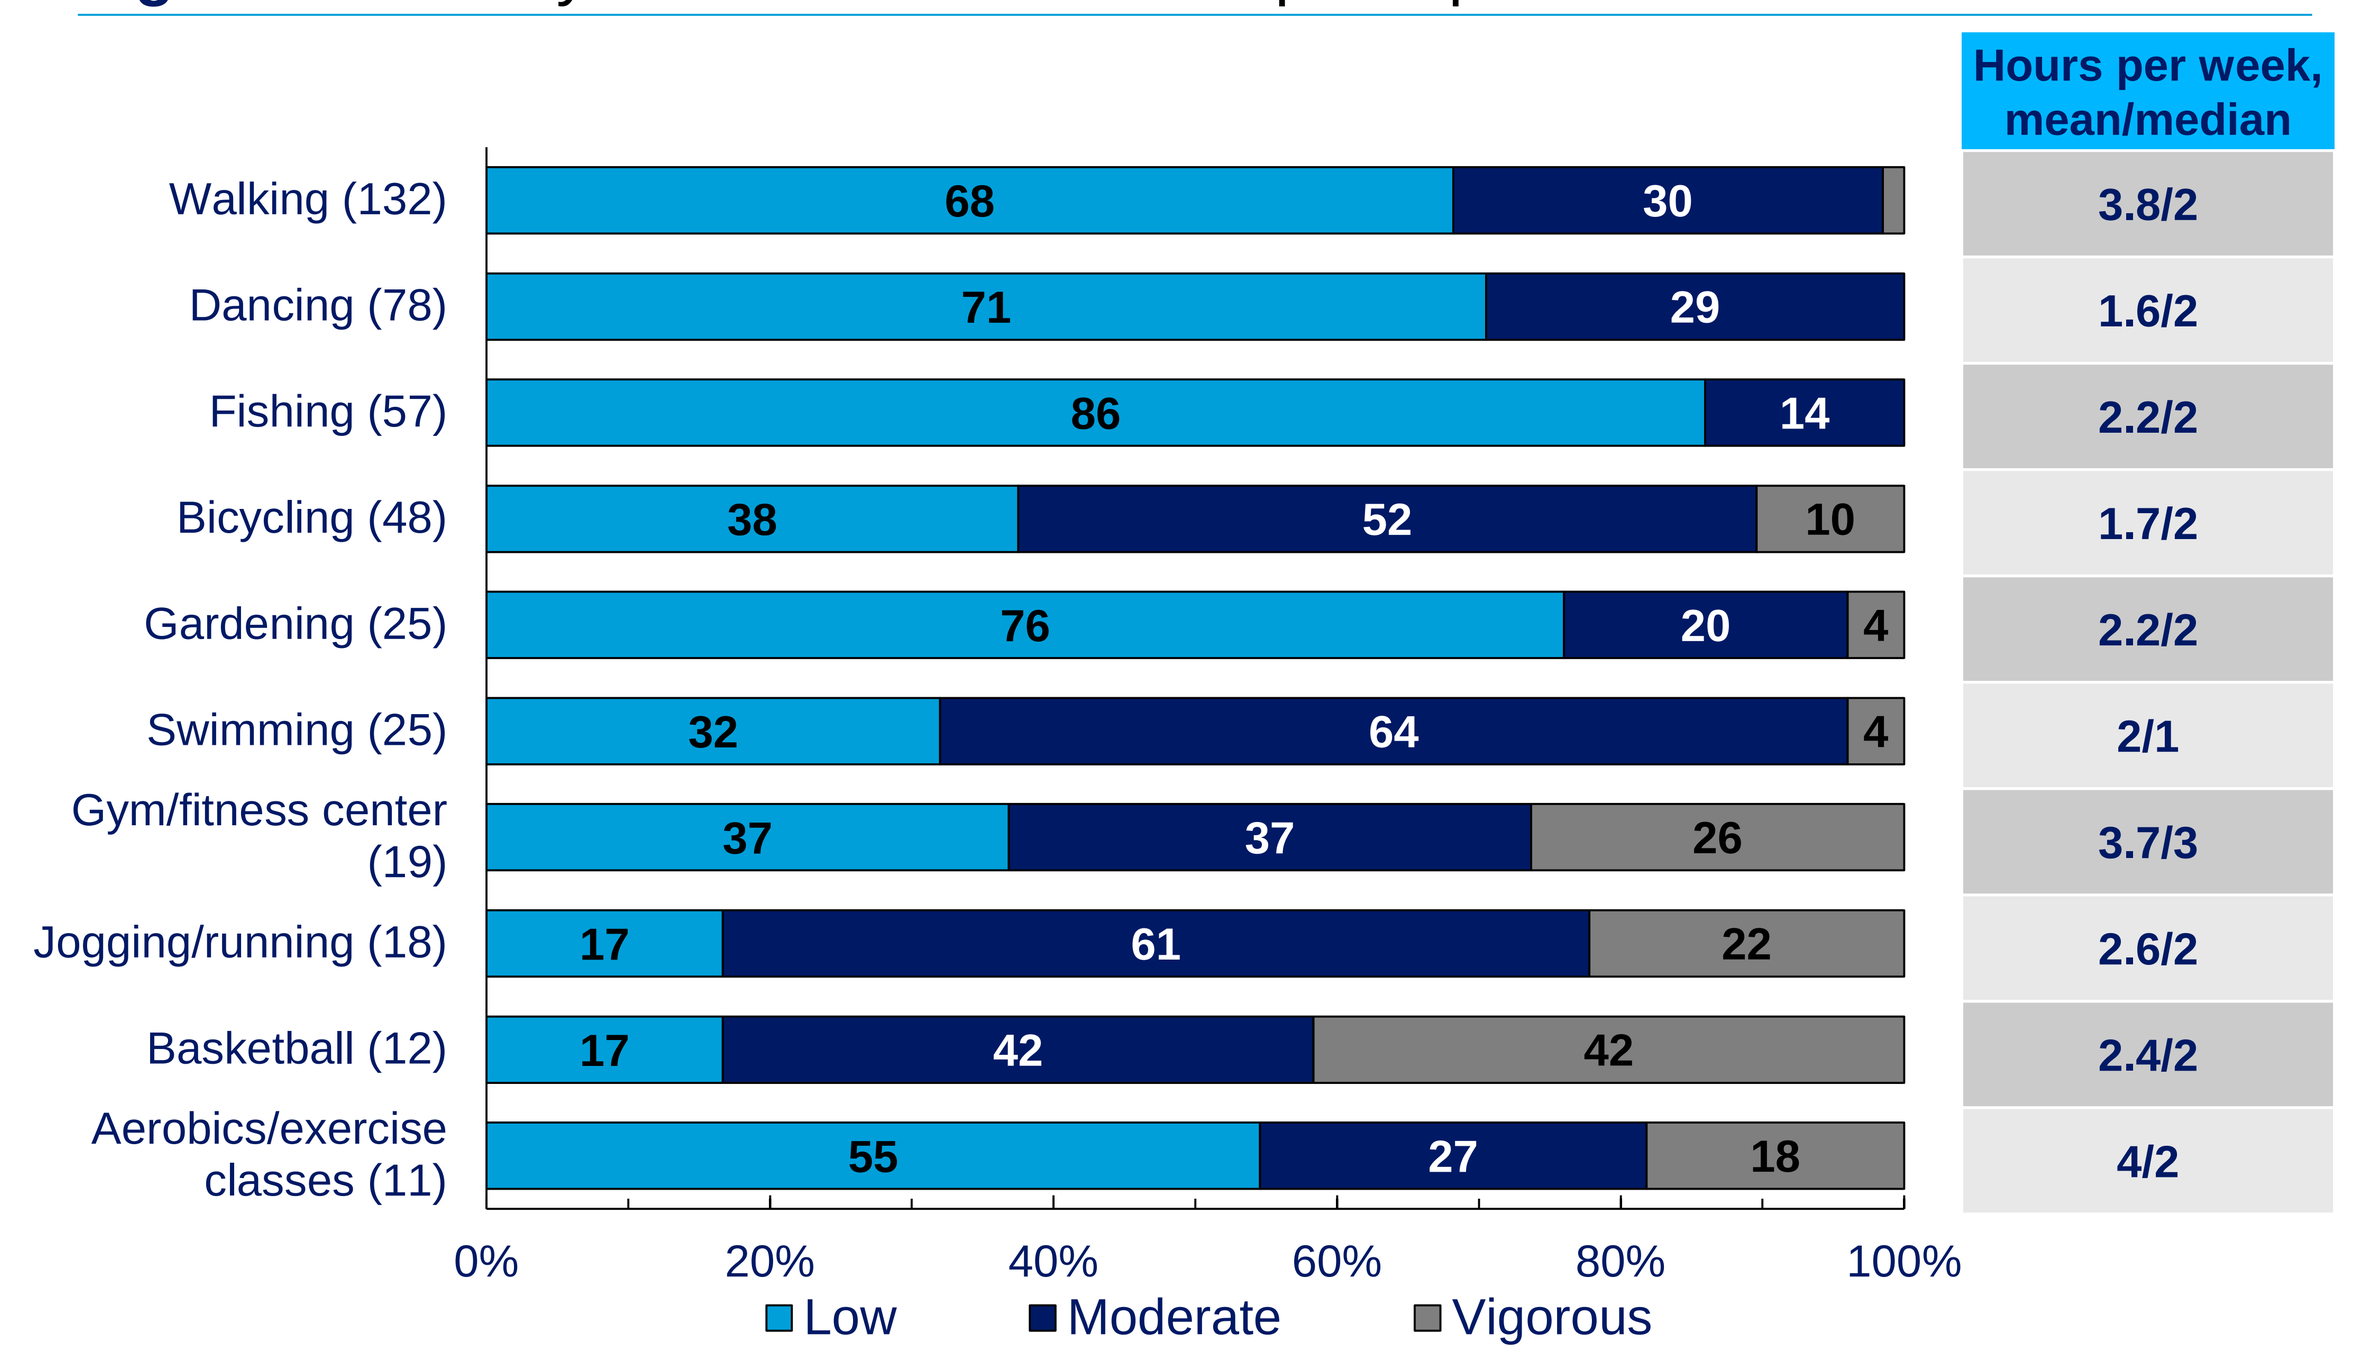

Hours per week, mean/median
Walking (132) 3.8/2
Dancing (78) 1.6/2
Fishing (57) 2.2/2
Bicycling (48) 1.7/2
Gardening (25) 2.2/2
Swimming (25) 2/1
Gym/fitness center (19) 3.7/3
Jogging/running (18) 2.6/2
Basketball (12) 2.4/2
Aerobics/exercise classes (11) 4/2
Low
Moderate
Vigorous

Parentheses indicate the numbers of PWH in each category.

- The most common activities that had been discontinued (which participants reported no longer being able to perform or considered too risky to continue) were bicycling (16%), swimming (12%), dancing (10%), jogging/running (7%), and basketball (6%).
- The top 5 desired activities in which PWH were unable to participate because of their hemophilia were basketball (27%), football (12%), skiing (9%), soccer (8%), and surfing (8%).
- Participants most commonly refrained from participating in activities because of fear and/or risk of bruising and muscle/joint bleeding (**Figure 3**).

Figure 3 Reasons for not participating in activities

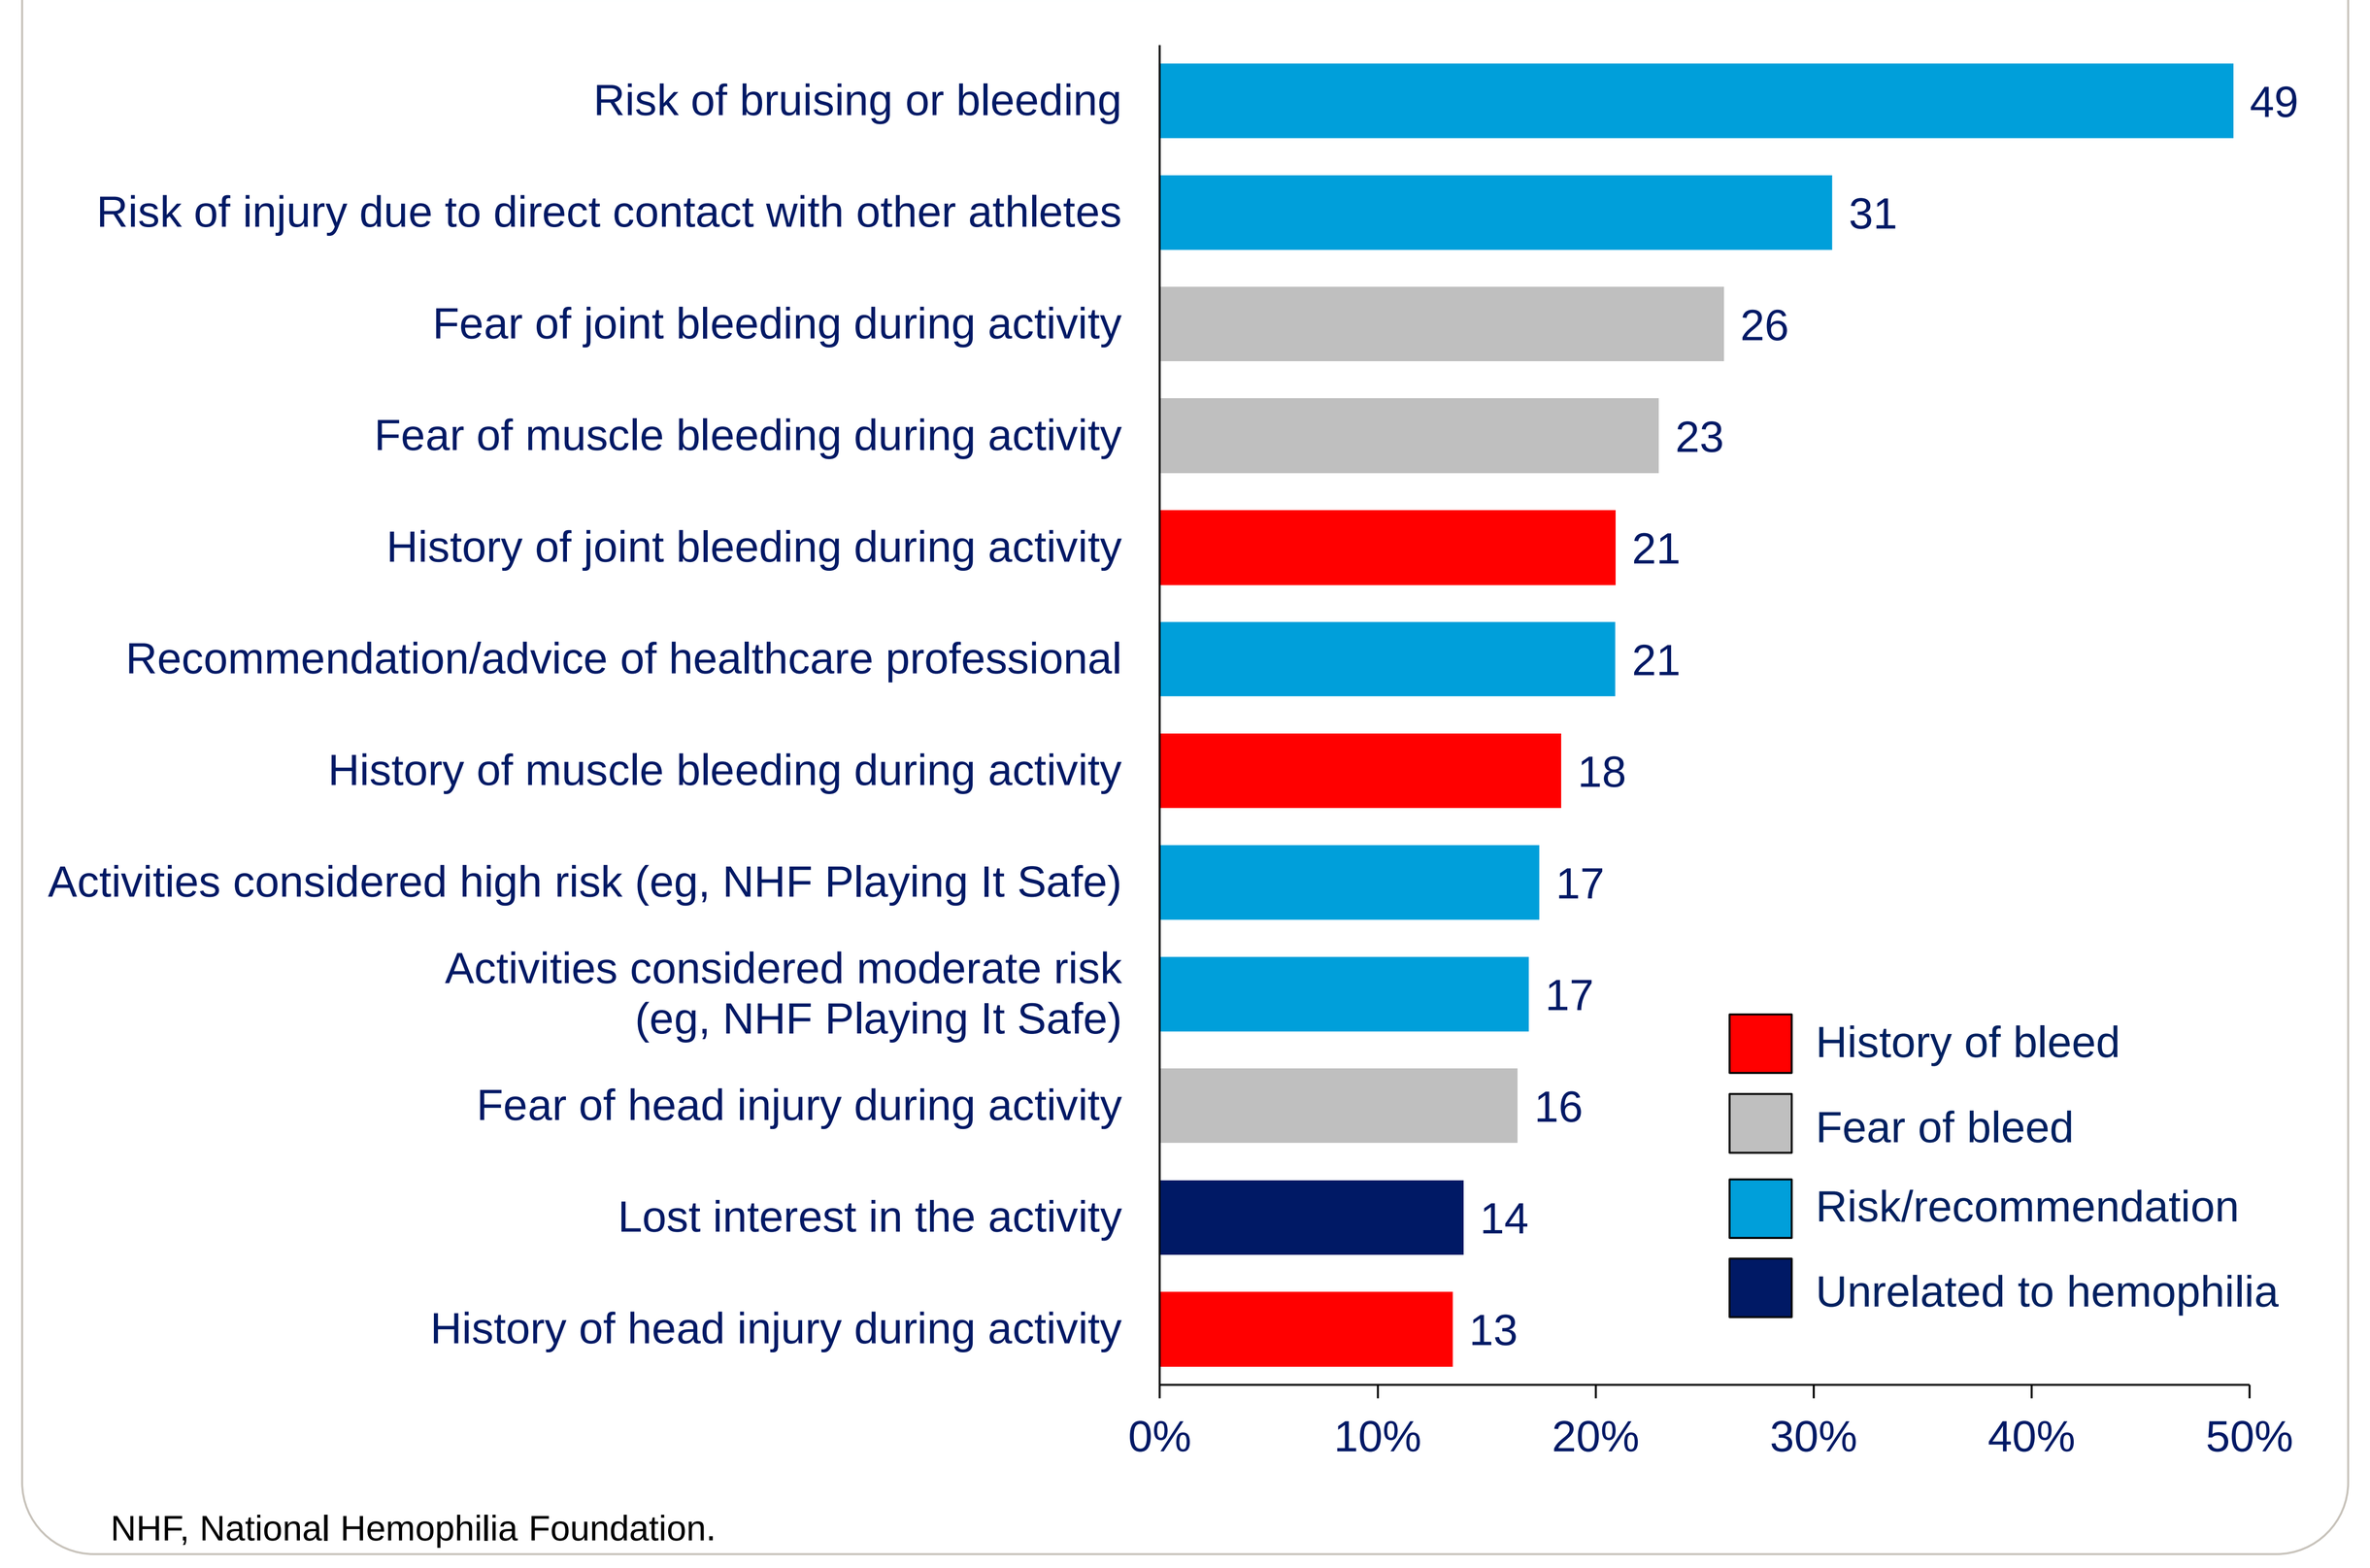

Risk of bruising or bleeding 49
Risk of injury due to direct contact with other athletes 31
Fear of joint bleeding during activity 26
Fear of muscle bleeding during activity 23
History of joint bleeding during activity 21
Recommendation/advice of healthcare professional 21
History of muscle bleeding during activity 18
Activities considered high risk (eg, NHF Playing It Safe) 17
Activities considered moderate risk (eg, NHF Playing It Safe) 17
Fear of head injury during activity 16
Lost interest in the activity 14
History of head injury during activity 13
History of bleed
Fear of bleed
Risk/recommendation
Unrelated to hemophilia

NHF, National Hemophilia Foundation.

Participation in activities across life stages

- Rates of participation in selected common current and high-risk activities are shown in **Figure 4**.
 - Participation in relatively low-risk activities (walking and dancing) increased from young ages through adulthood.
 - Current or past participation in higher-risk activities generally peaked during middle school or high school, a stage when contact and injury risk are higher and sports tend to become year-round.

Figure 4 Levels of participation in selected activities across life stages

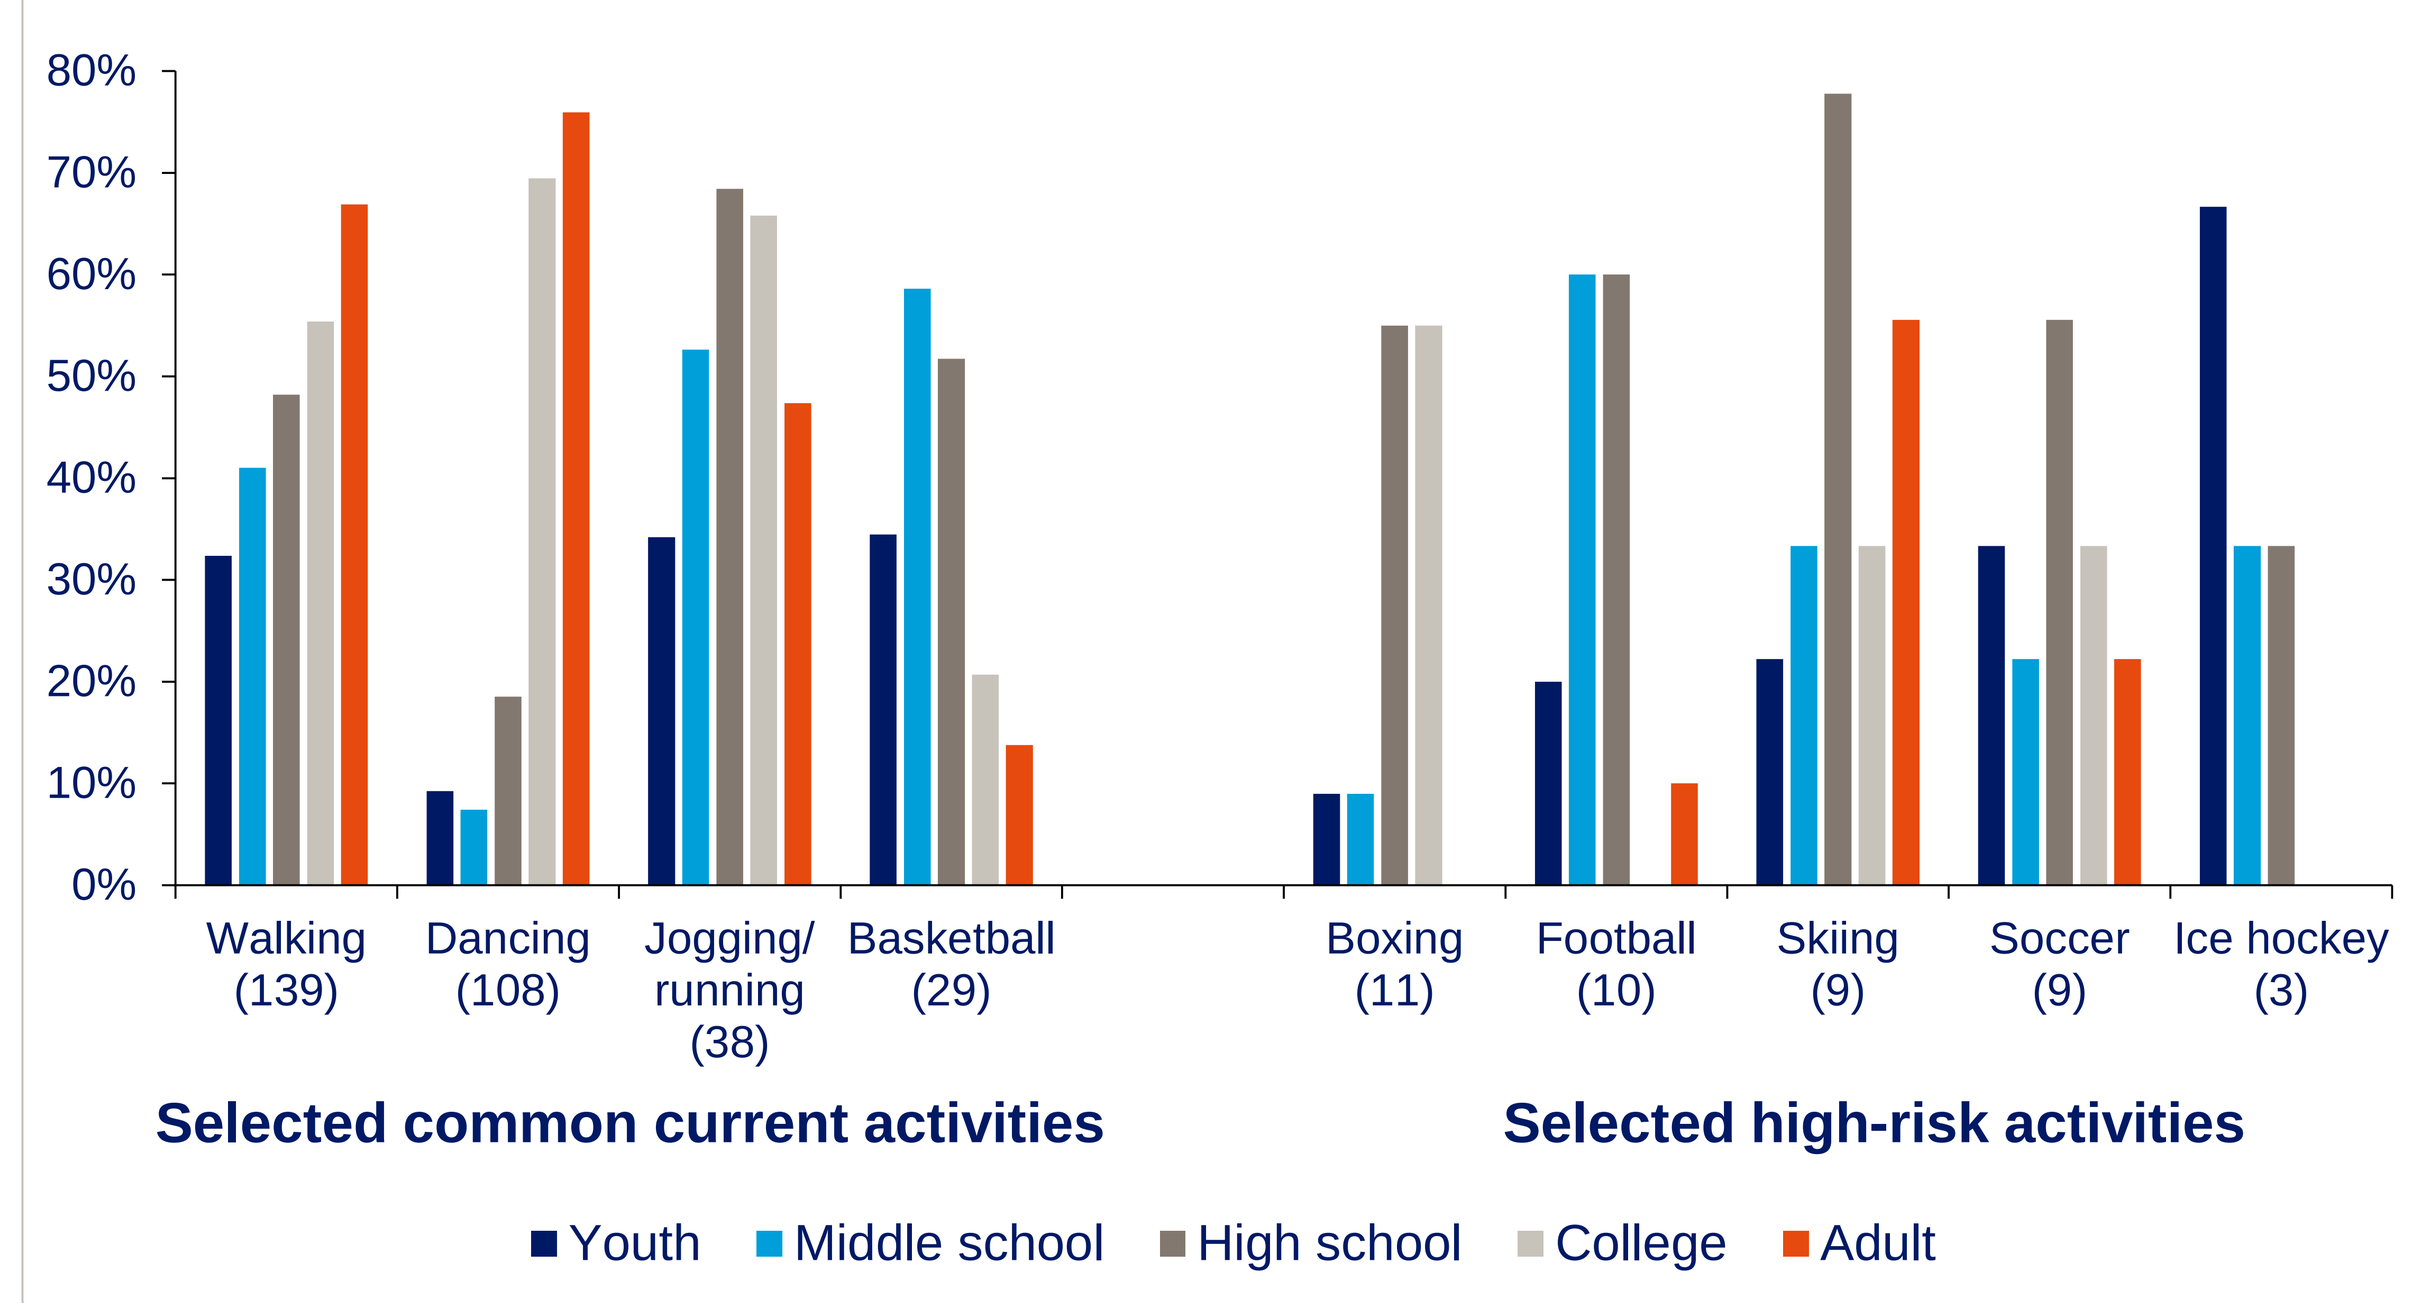

Walking (139)
Dancing (108)
Jogging/ running (38)
Basketball (29)
Boxing (11)
Football (10)
Skiing (9)
Soccer (9)
Ice hockey (3)
Selected common current activities
Selected high-risk activities
Youth
Middle school
High school
College
Adult

Parentheses indicate the numbers of PWH in each category.

Changes to treatment around activity

- Most PWH reported making significant or moderate changes in their treatment regimens to accommodate activities (**Figure 5**).
 - Higher numbers of participants with severe hemophilia, compared with those with mild or moderate hemophilia, reported significant changes.
 - PWH receiving routine infusions were more likely than those receiving on-demand treatment to make significant or moderate changes.

References

1. Forsyth AL et al. *Patient Prefer Adherence.* 2015;9:1549-1560.
2. Witkop M et al. *Am J Hematol.* 2015;90(suppl 2):S3-S10.

Conclusions

- Hemophilia affected engagement in activities for nearly all participants, regardless of gender or hemophilia severity.
- Adults with even mild/moderate hemophilia discontinued activities that were generally of higher risk; most activities were reduced by the time of college.
- Reluctance to continue participating in activities was mainly due to risk/fear of joint/muscle bleeding or contact-related bleeding; many PWH reported a history of activity-associated bleeds.
- PWH frequently adjusted their treatment regimen in order to participate in activities.

Figure 5 Changes to treatment regimen around activity, overall (A) and by hemophilia severity and treatment regimen (B)

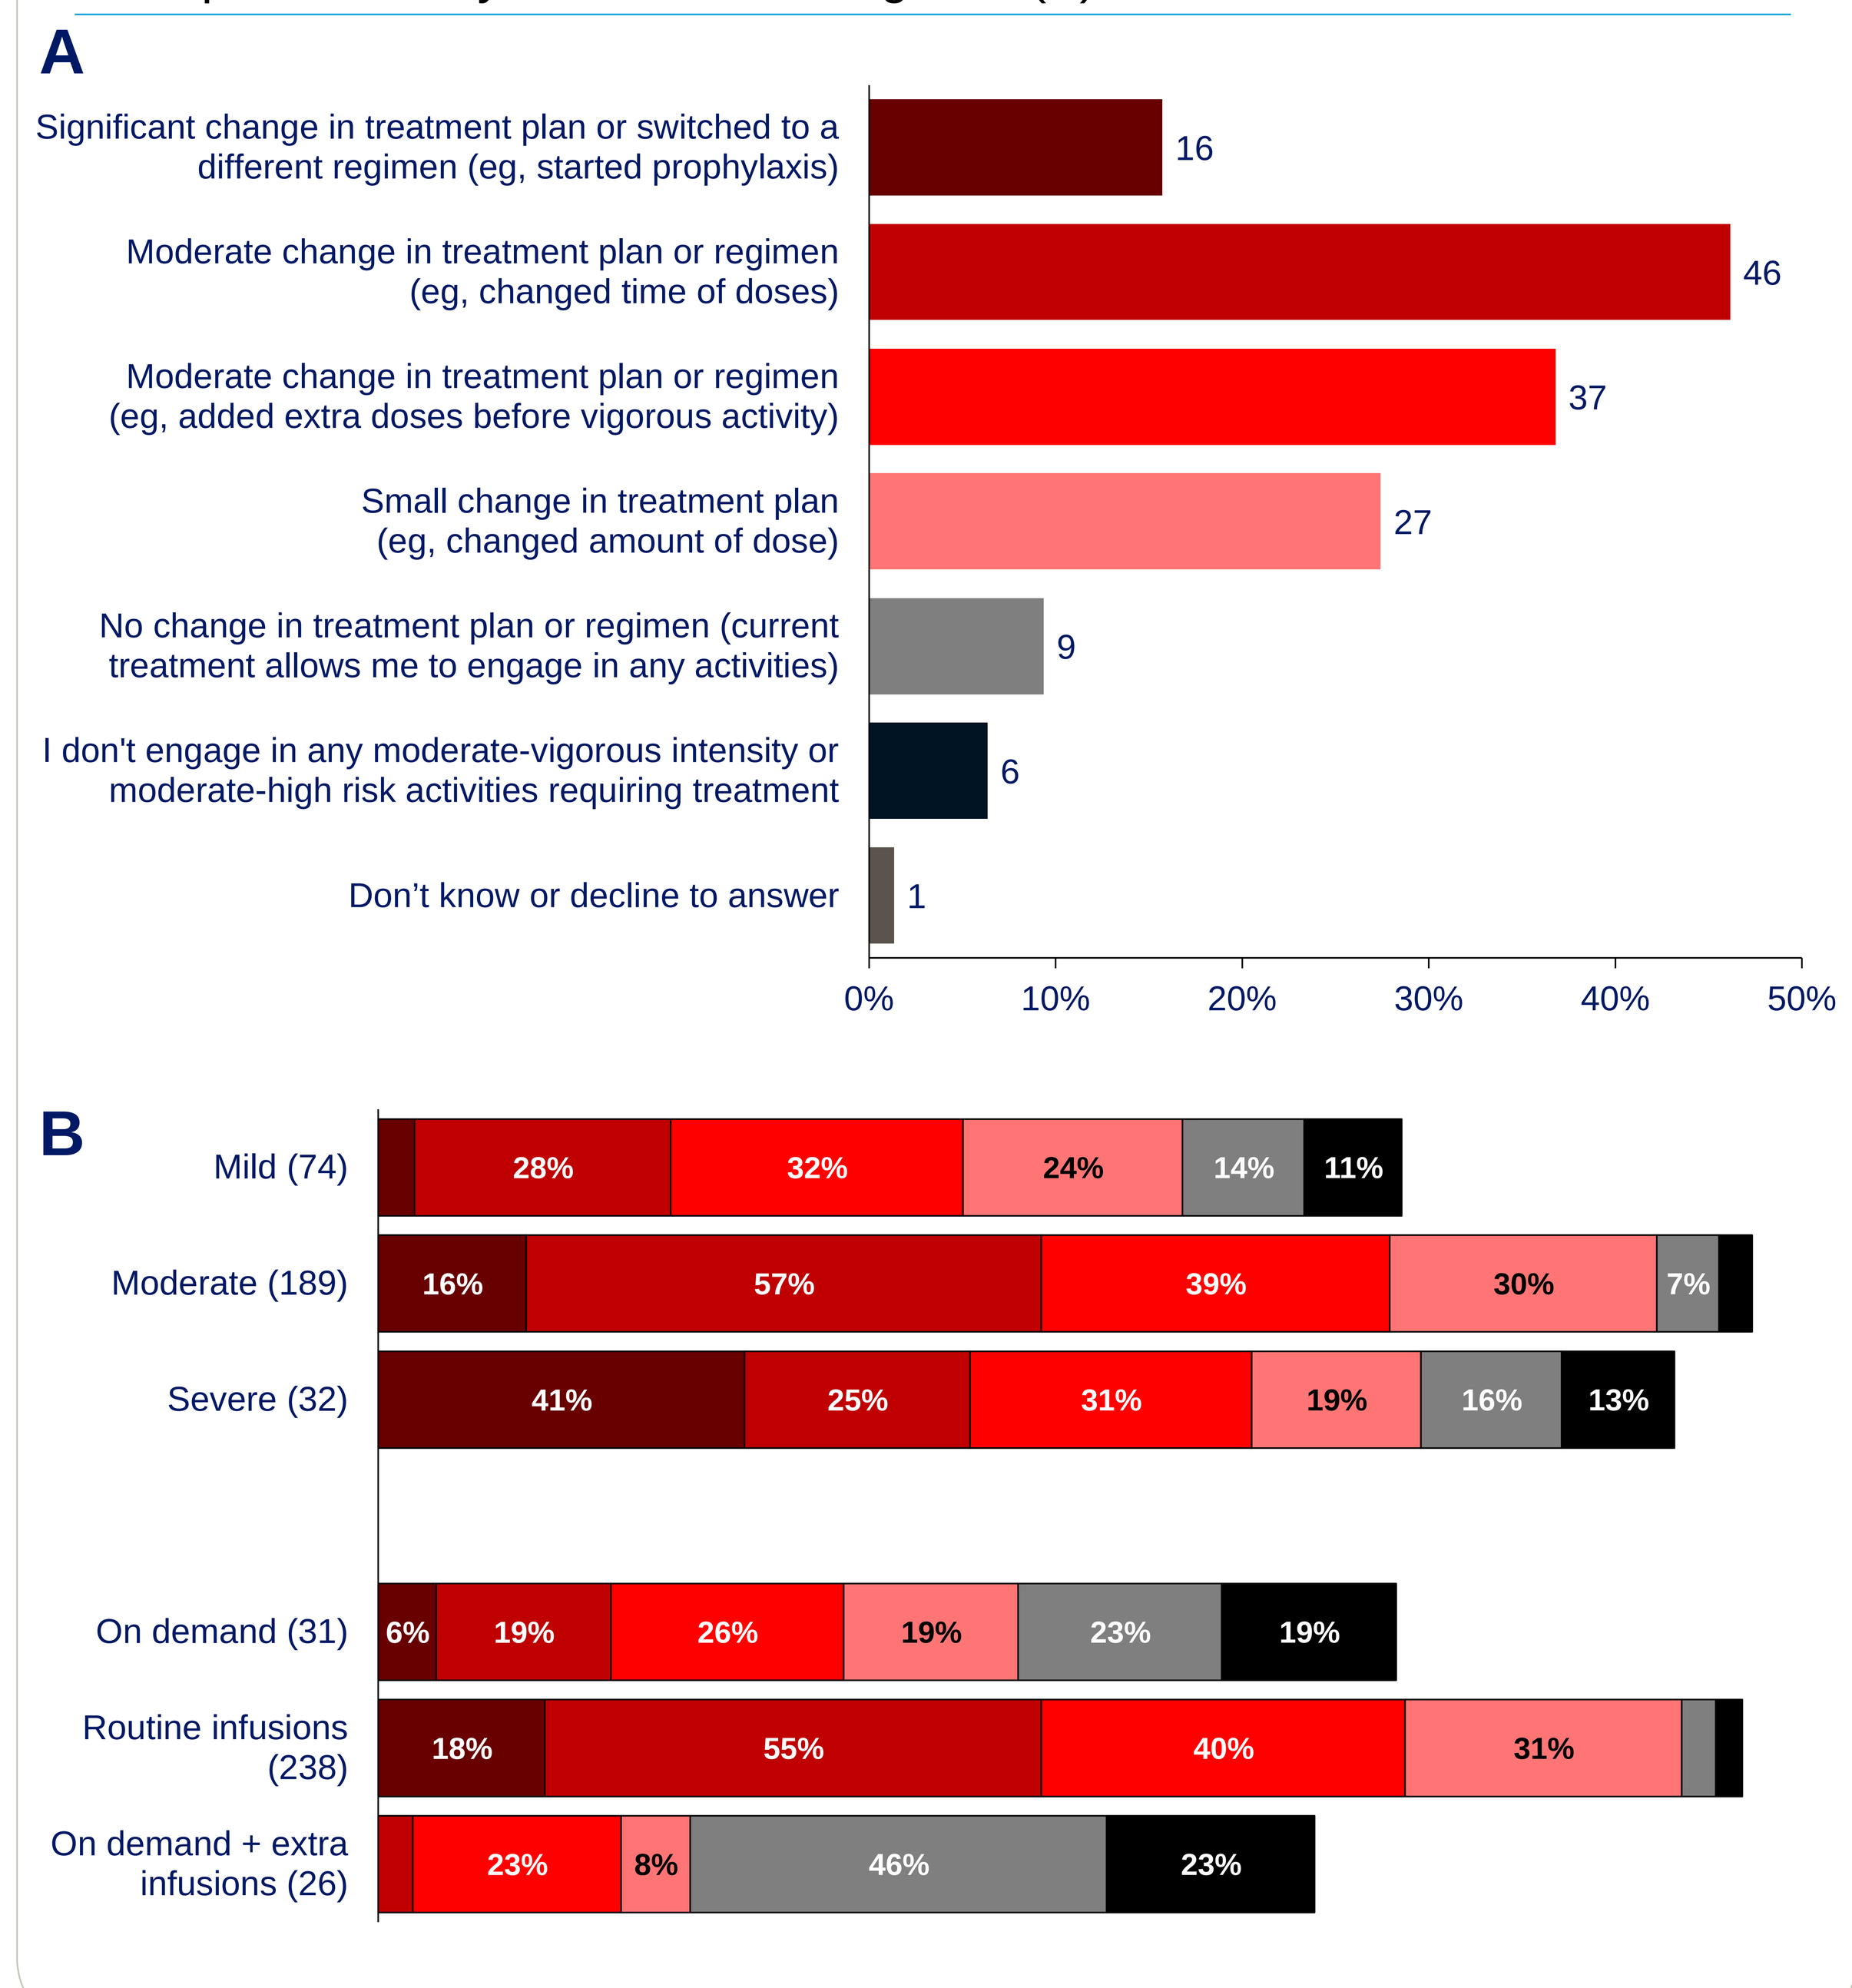

A
Significant change in treatment plan or switched to a different regimen (eg, started prophylaxis) 16
Moderate change in treatment plan or regimen (eg, changed time of doses) 46
Moderate change in treatment plan or regimen (eg, added extra doses before vigorous activity) 37
Small change in treatment plan (eg, changed amount of dose) 27
No change in treatment plan or regimen (current treatment allows me to engage in any activities) 9
I don't engage in any moderate-vigorous intensity or moderate-high risk activities requiring treatment 6
Don't know or decline to answer 1
B
Mild (74)
Moderate (189)
Severe (32)
On demand (31)
Routine infusions (238)
On demand + extra infusions (26)

Parentheses indicate the numbers of PWH in each category. Multiple selections were allowed.

Conflict of interest disclosure

KB has served as a consultant for Bayer and Novo Nordisk and as a speaker for Baxter, Bayer, and Novo Nordisk. GH has received grant/research support from Bayer and Novo Nordisk and served on advisory boards for Biogen Idec and on the speakers bureau for Emergent BioSolutions. MW is a speaker for Novo Nordisk and has participated in their advisory boards. SP has served as a consultant for Novo Nordisk and has given a talk for Baxalta. SD has nothing to declare. SC has received honoraria from Novo Nordisk and Solutions Sights. NF has nothing to declare. KH is an employee of the Hemophilia Federation of America. NI and DC are employees of Novo Nordisk Inc.

This trial was sponsored by Novo Nordisk Inc. and is registered with ClinicalTrials.gov (NCT02568202).
The authors acknowledge the medical writing assistance of Kathy Carpenter.
Presented at the World Federation of Hemophilia (WFH) 2016 World Congress, July 24-28, 2016, Orlando, FL, USA.

Poster Presented at:
DOI: 10.3252/pso.eu.WFH2016.2016
Sports and Bleeding Disorders
Kim Baumann
174--PP-T
WFH2016