HEMOPHILIA CARE IN NATIONAL INSTITUTE OF HEMATOLOGY AND BLOOD TRANSFUSION

48

Nguyen Thi Mai, Nguyen Anh Tri, Bach Quoc Khanh
(National Institute of Hematology and Blood Transfusion, Hanoi, Vietnam). Email: hemophiliavn@gmail.com

INTRODUCTION:

Hemophilia is a X linked bleeding disorder caused by deficiency of factor VIII/IX. In Vietnam, estimated, there are 6,000 patients with hemophilia, 30,000 carriers, in which only 40% is diagnosed and managed. Currently, we have 7 hospitals give care for hemophilia patient, among of which, the National Institute of Hematology and Blood Transfusion (NIHBT) is the biggest centre, which manage over 1,300 patients with hemophilia and other congenital bleeding disorders.

RESULTS:

The patients were treated on demand and also comprehensive cared by a trained team, was screened blood transfusion transmitted diseases and inhibitors periodically, were counseled to living with the disease as well as genetic counseling.

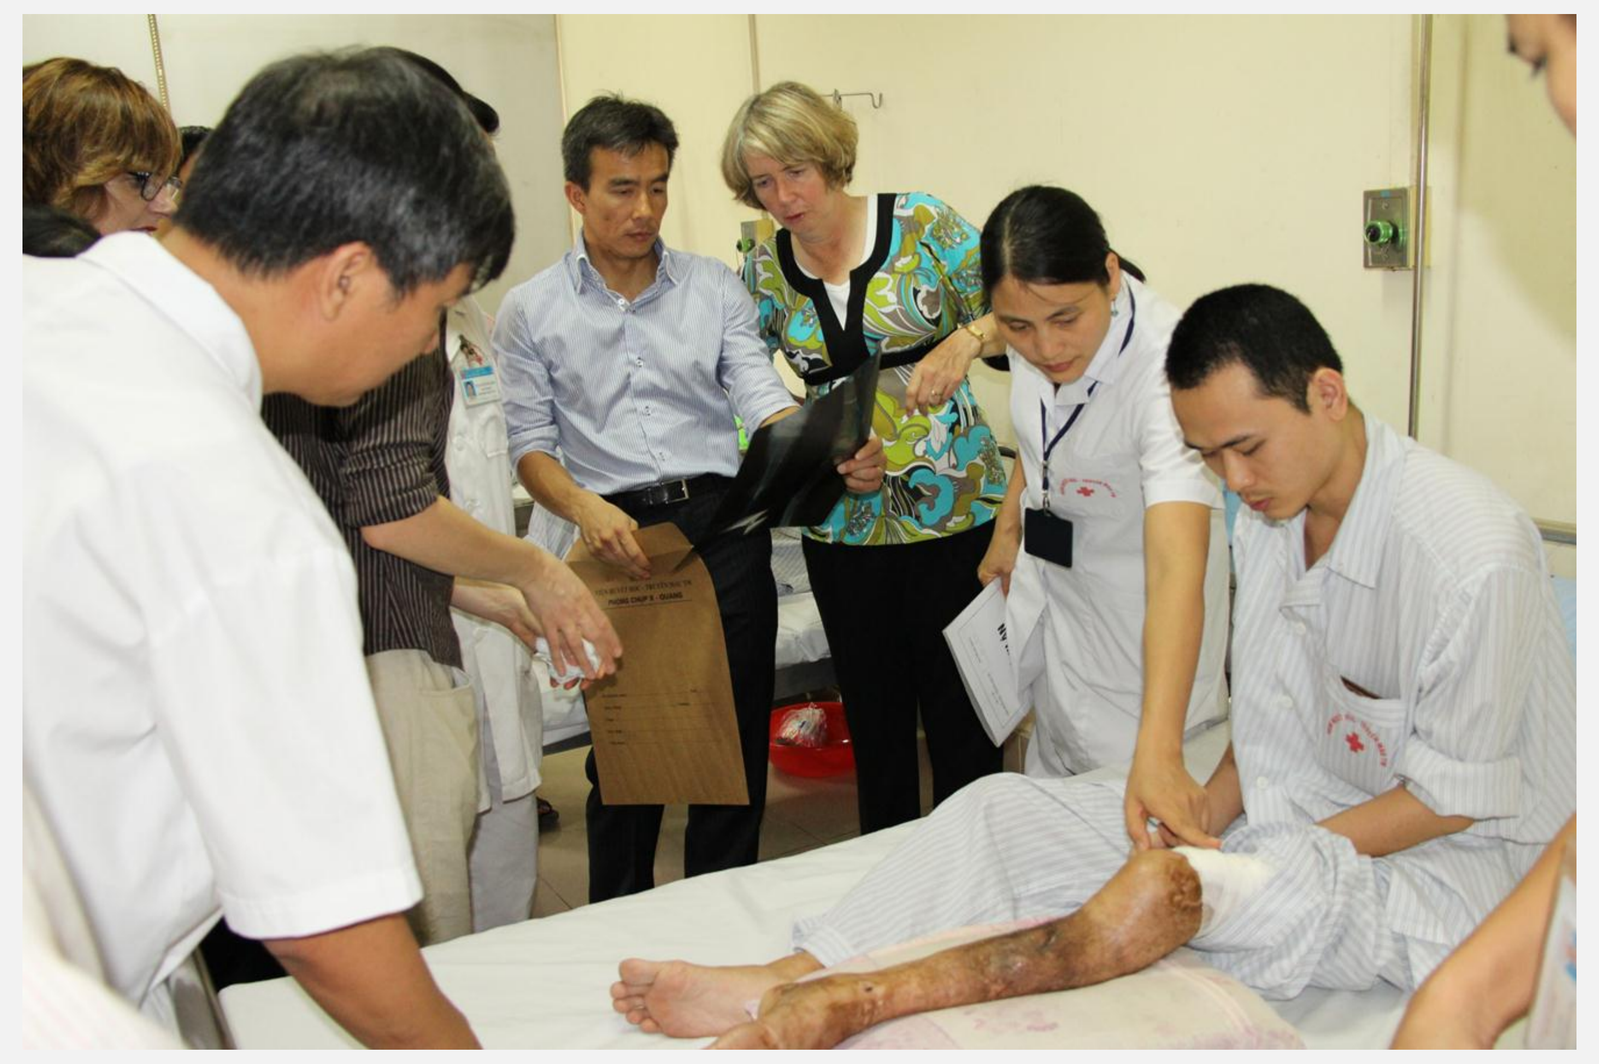

Comprehensive care for patient with hemophilia

RESULTS (cont):

Among the hemophilia patients being managed in the NIHBT, there are 83% of hemophilia A, hemophilia B is 17%, 48.5% severity, 26.8% moderate, mild 24.7%.

Chart 1: Percentage of Hemophilia A and Hemophilia B patients in NIHBT

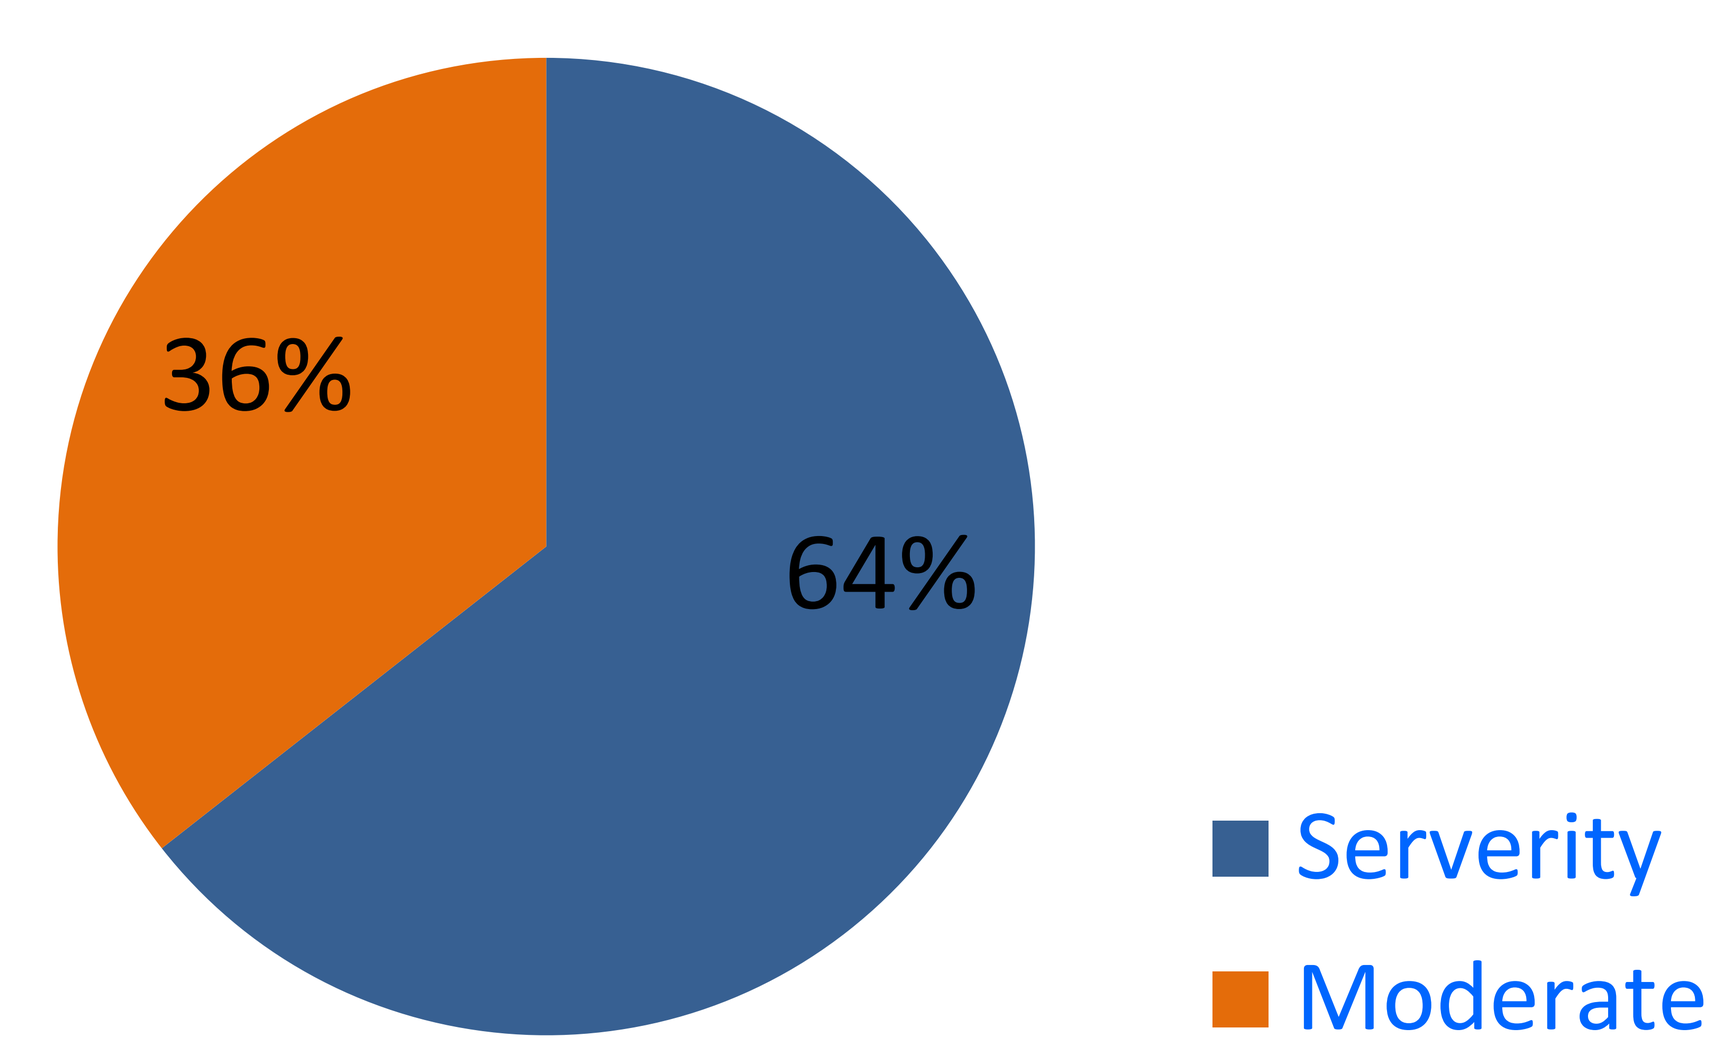
36%
64%
Serverity
Moderate

Chart 2: Percentage disease severity

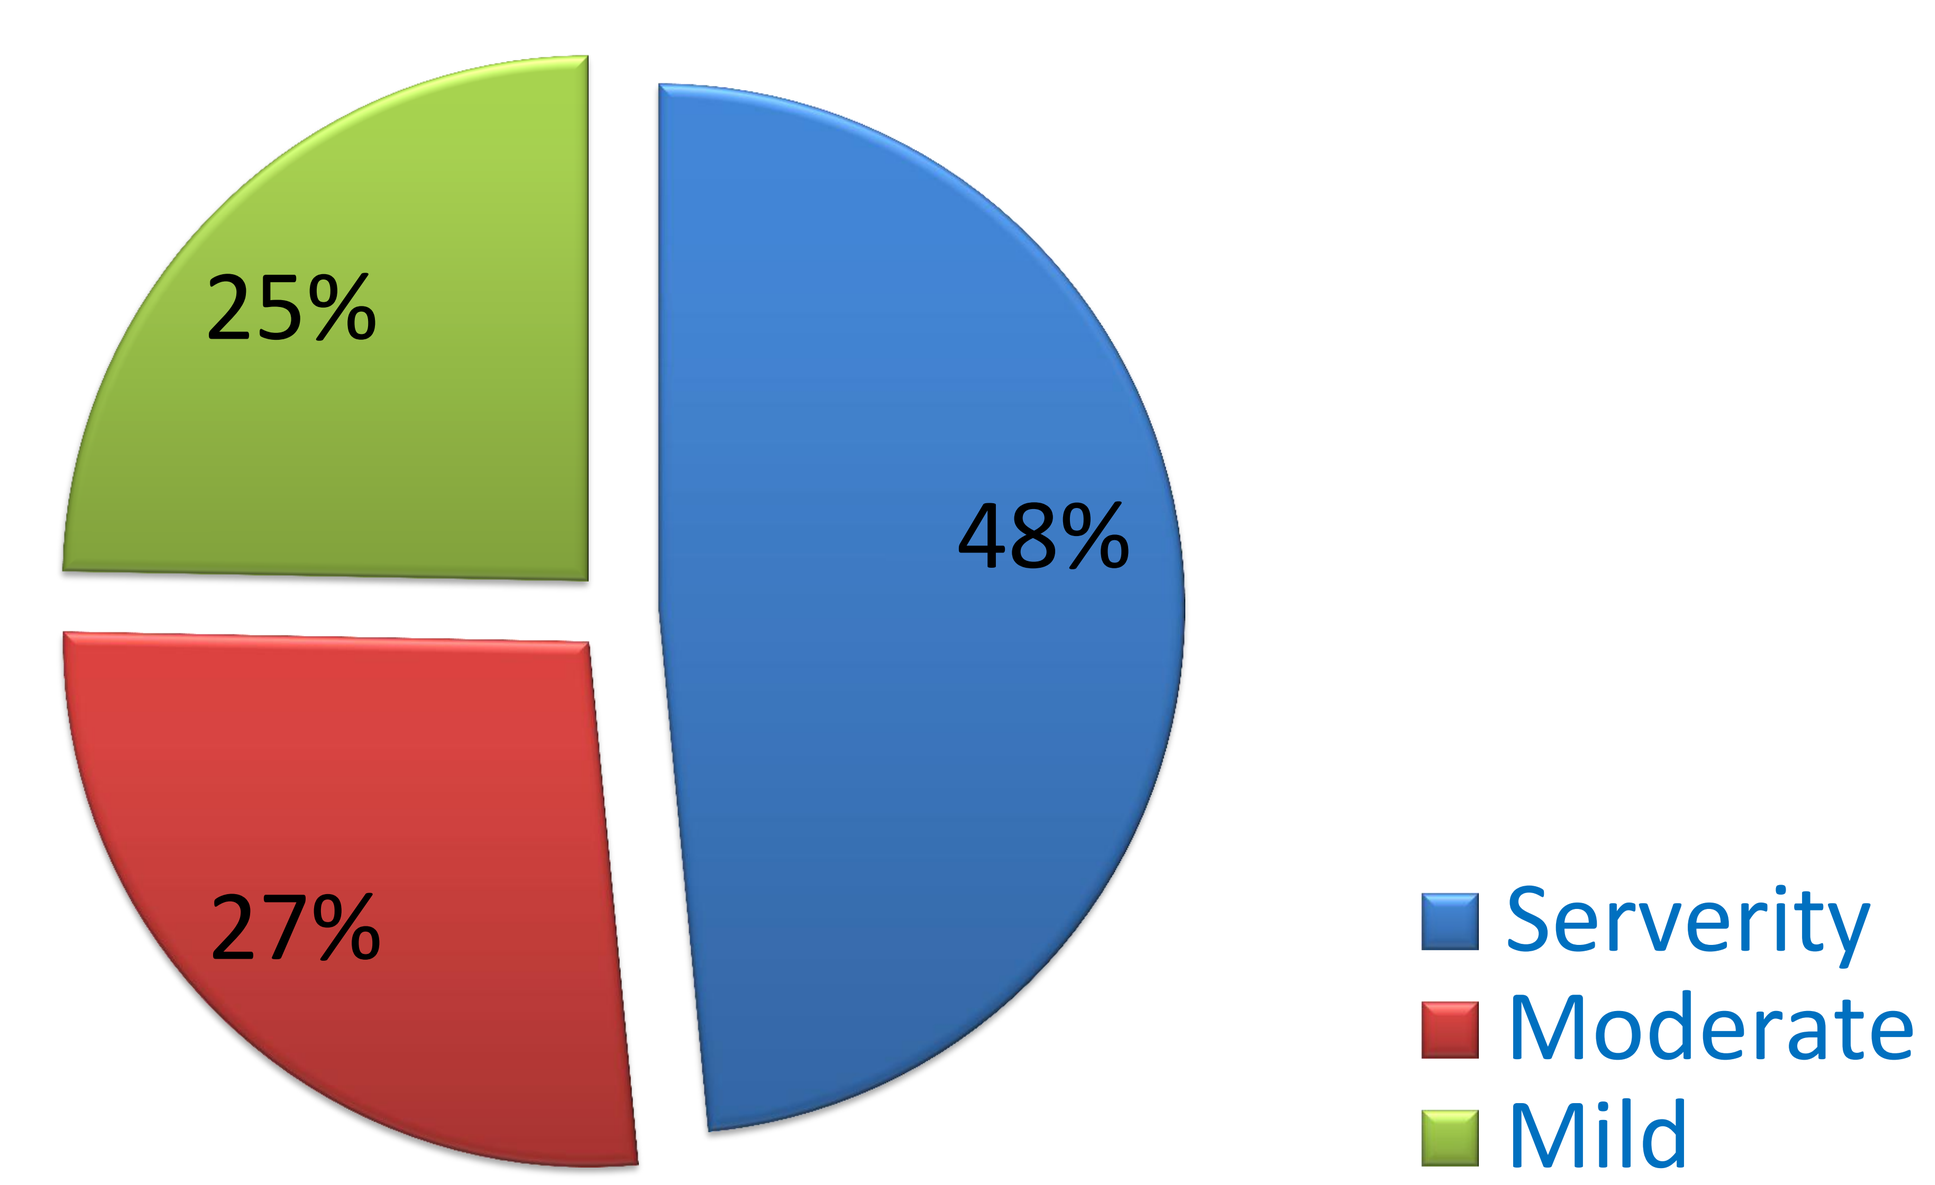
25%
48%
27%
Serverity
Moderate
Mild

In 2014, the Ministry of Health has issued "Guidelines for diagnosis and treatment of hemophilia". This is the basis to conduct a comprehensive treatment, early treatment of bleeding at home, low dose prophylaxis for severe hemophilia patients.

There are 77 patients (7.1%) positive with inhibitor, which most of them are hemophilia A. Currently, there are two types of blood products used for treatment as domestically produced products (cryoprecipitate, plasma) and imported products (plasma derived factor VIII/IX concentrates, recombinant factor VIIa).

The amount of factor concentrates used in recent years have tended to rise (national wide in 2014 FVIII used 0.1 ui/capita, FIX used 0.013ui/ capita), but still low compared with countries with similar income as well as considering the needs of the patient being treated.

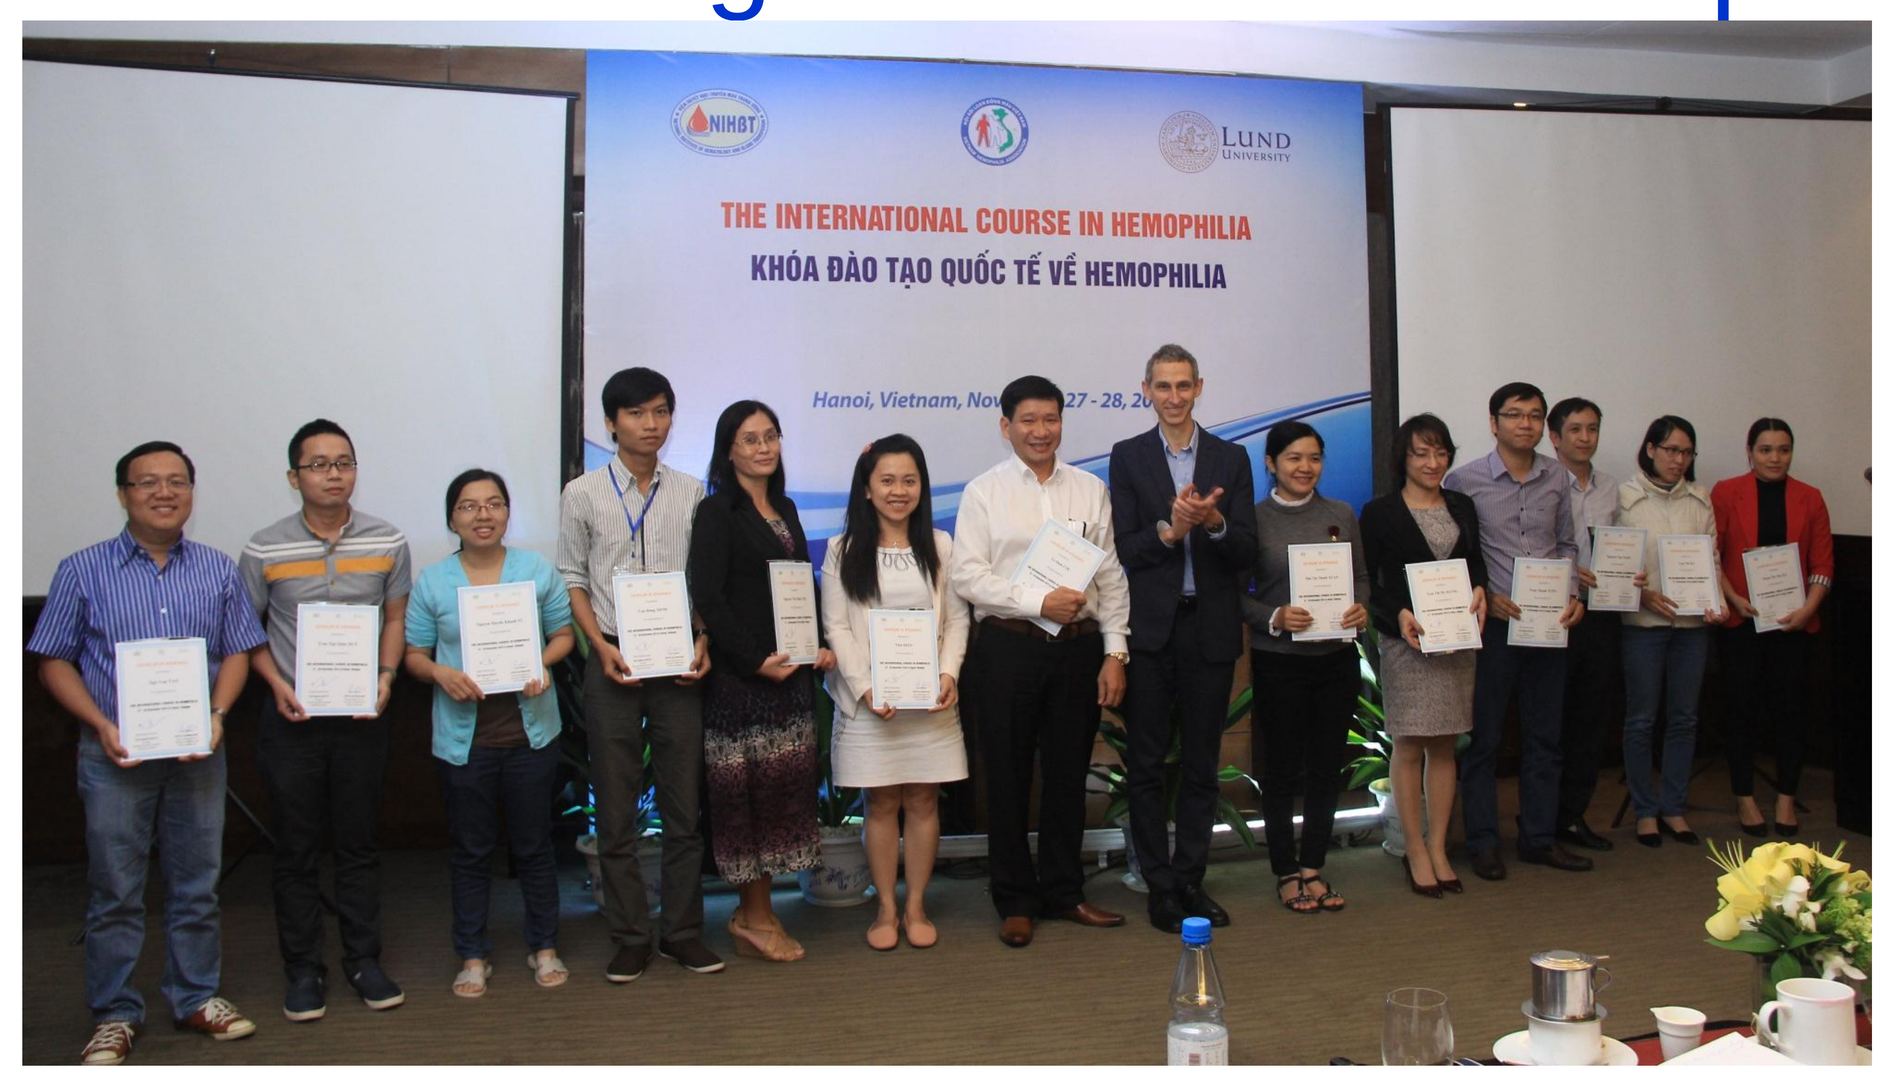
THE INTERNATIONAL COURSE IN HEMOPHILIA
KHÓA ĐÀO TẠO QUỐC TẾ VỀ HEMOPHILIA
LUND

International training course for doctors and medical staffs

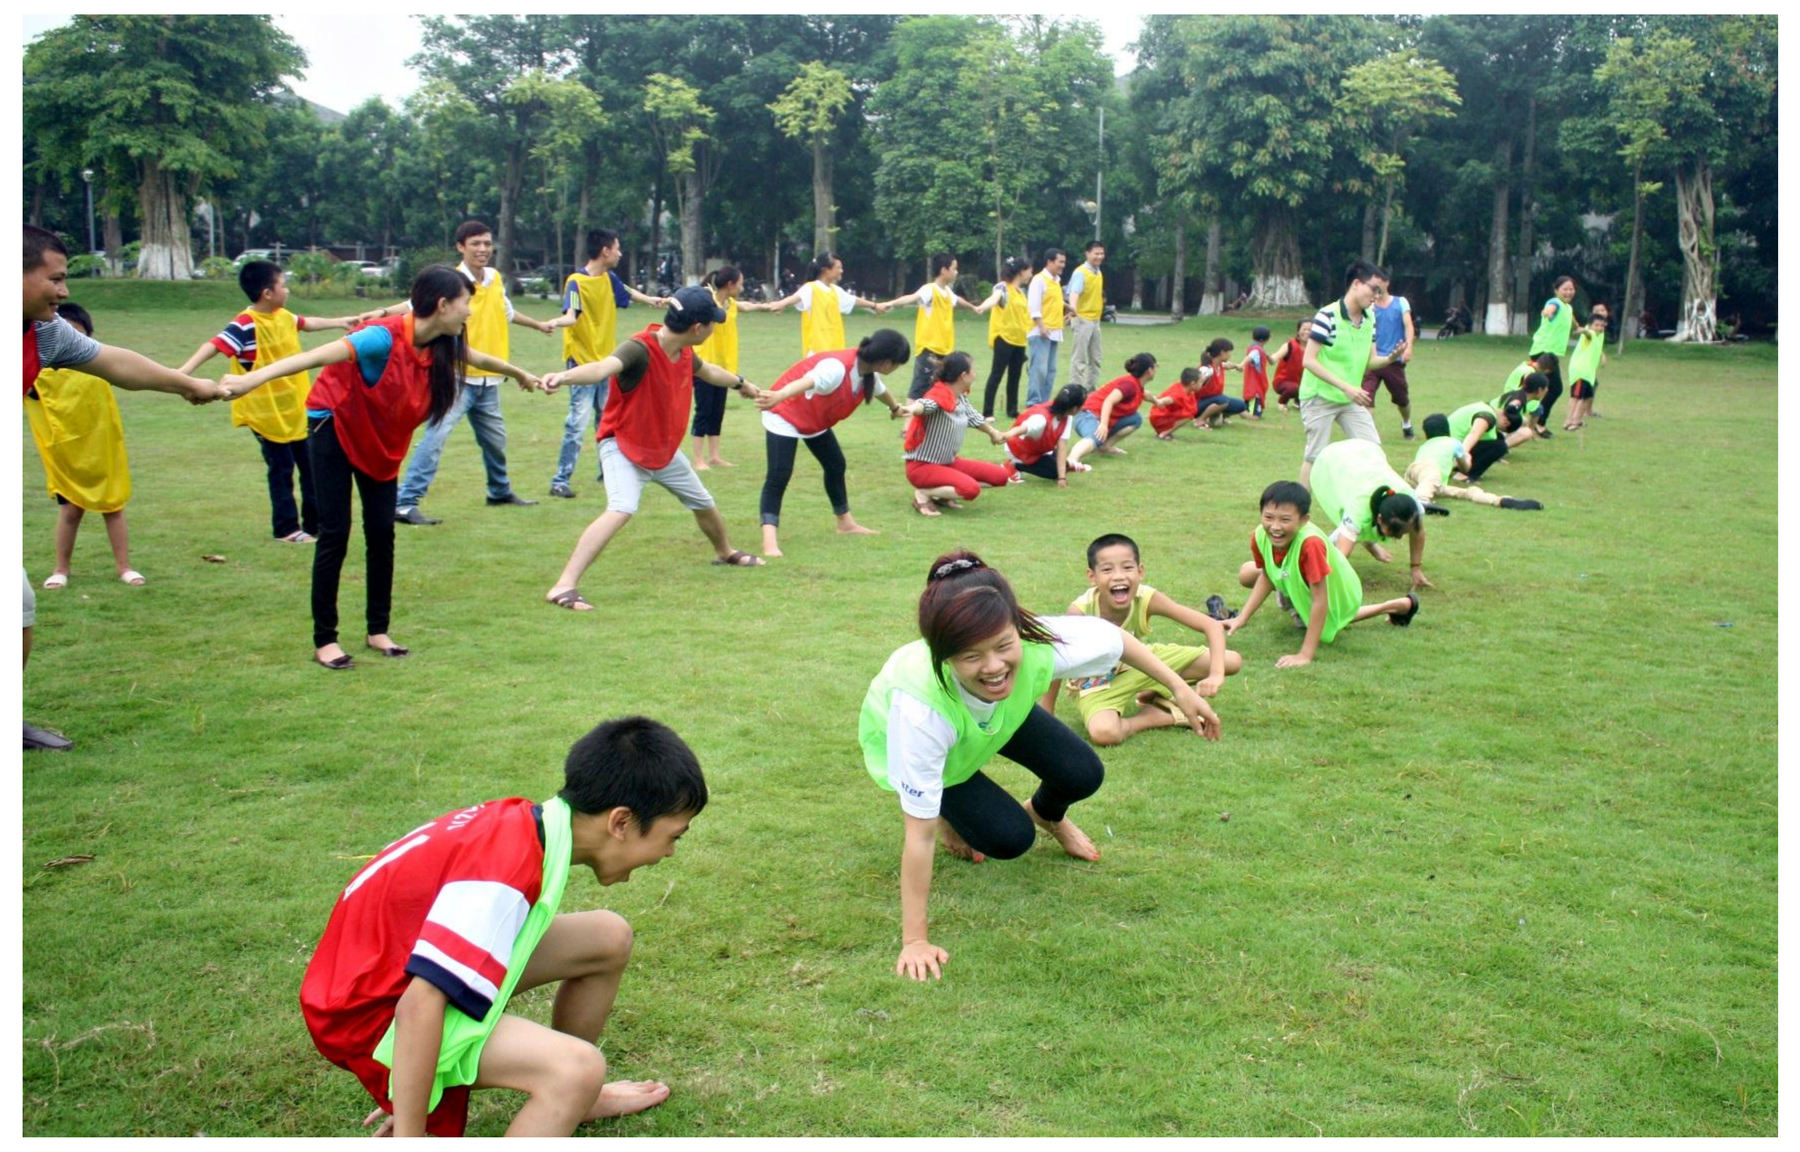

Summer camp for hemophilia patients in Vietnam

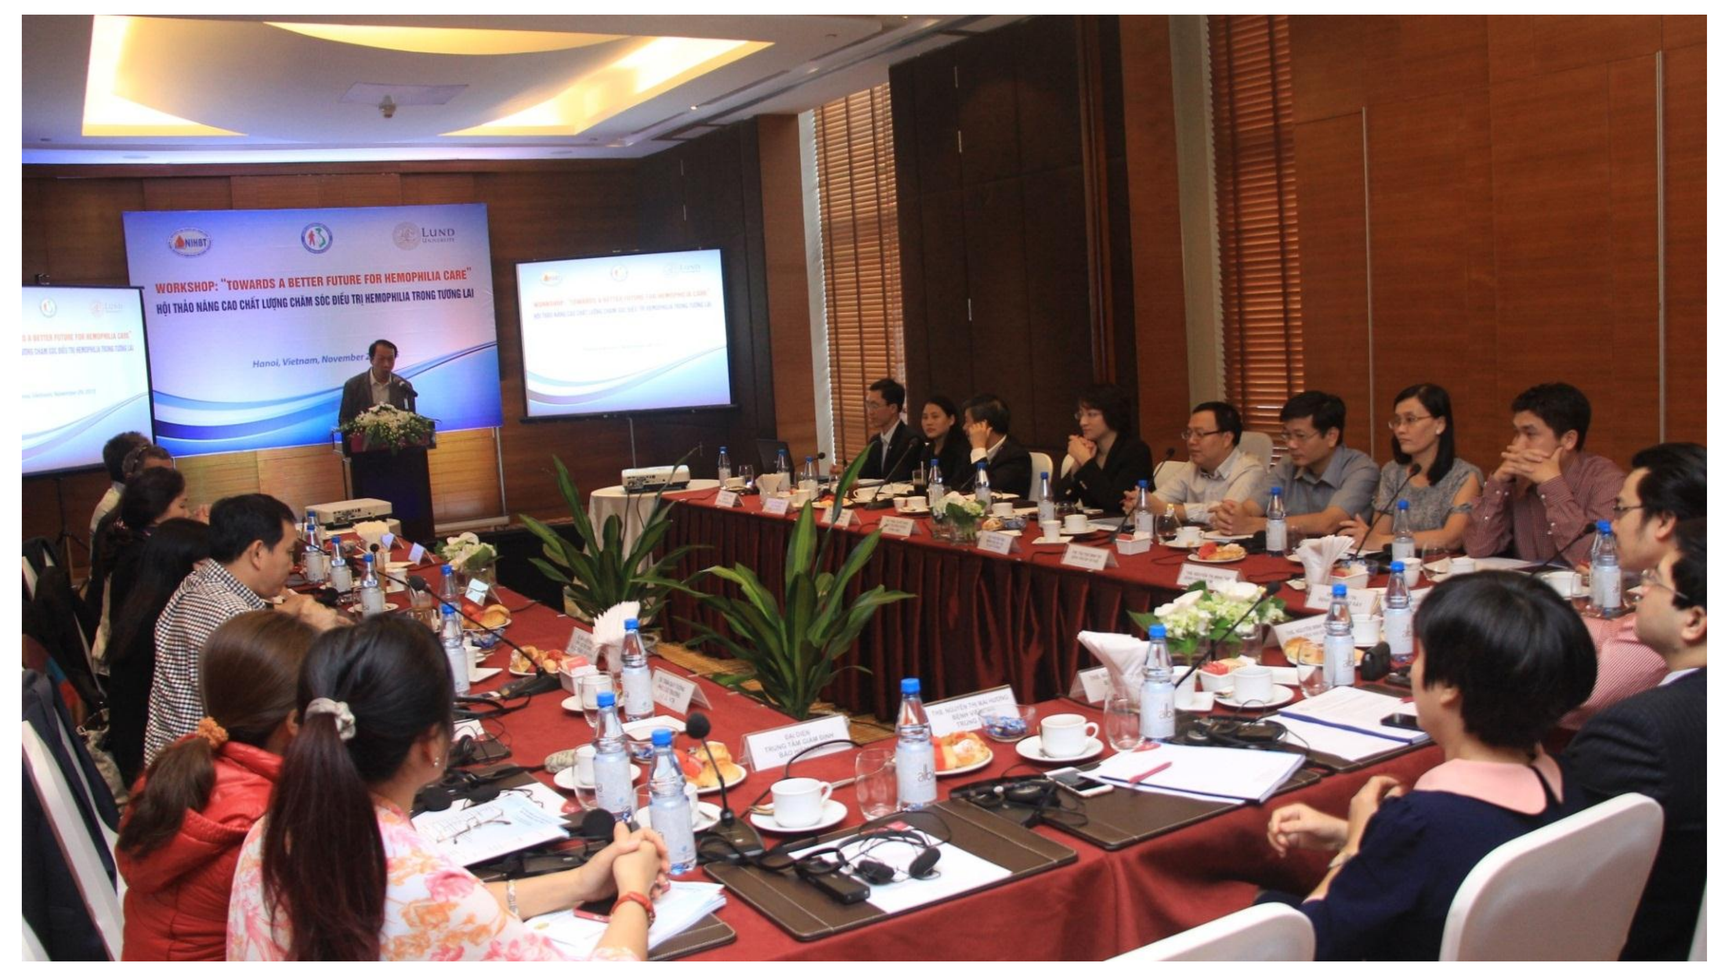

Advocacy with authorities to get better policies.

RESULTS (cont):

In addition, Hemophilia Centers of NIHBT is the core of the Vietnam Hemophilia Association (VHA). VHA has developed many activities such as education, training, advocacy, fundraising, humanitarian aide, international cooperation... to improve the quality of hemophilia care across the country.

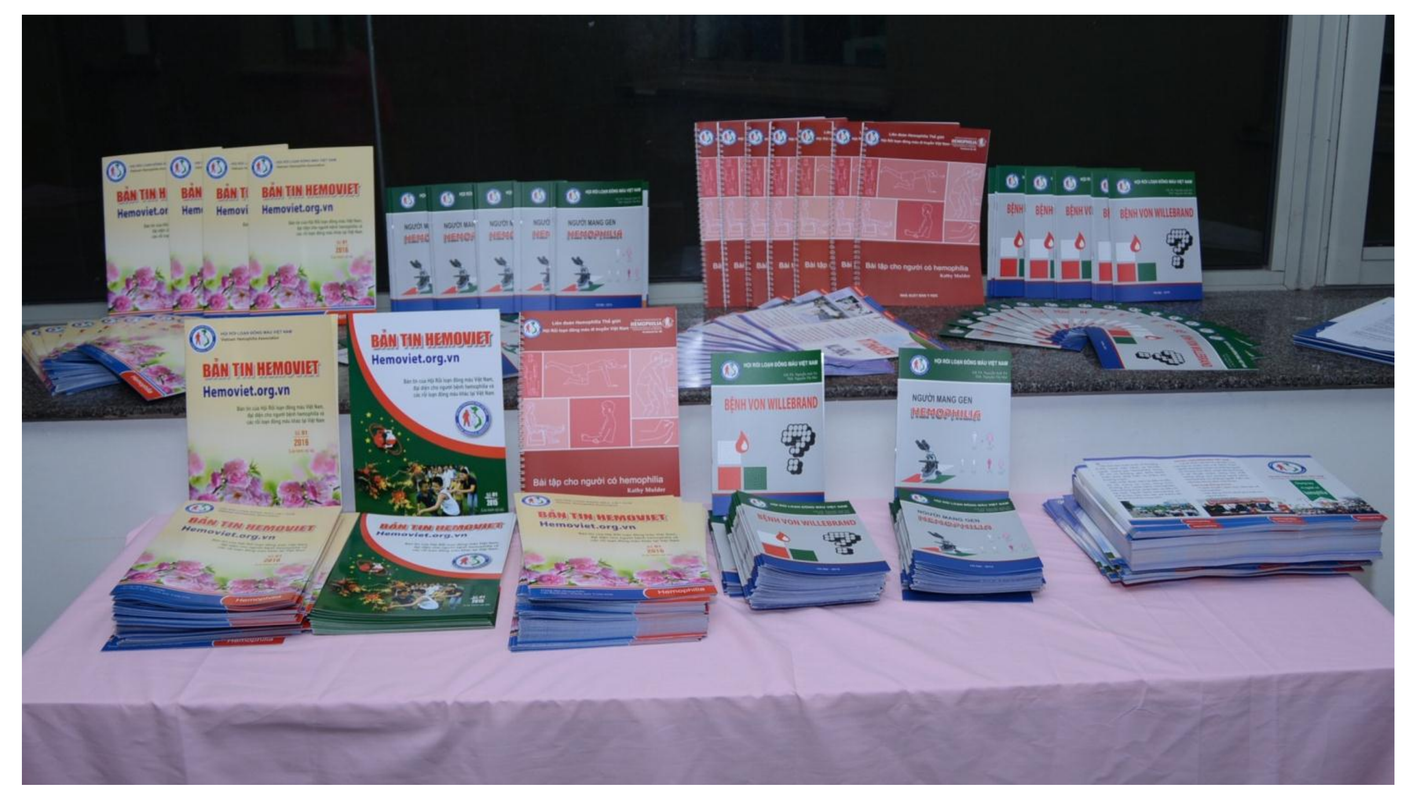

In 2012, both Hemophilia Center and the Society have won awards of World Federation of Hemophilia' Twinning program.

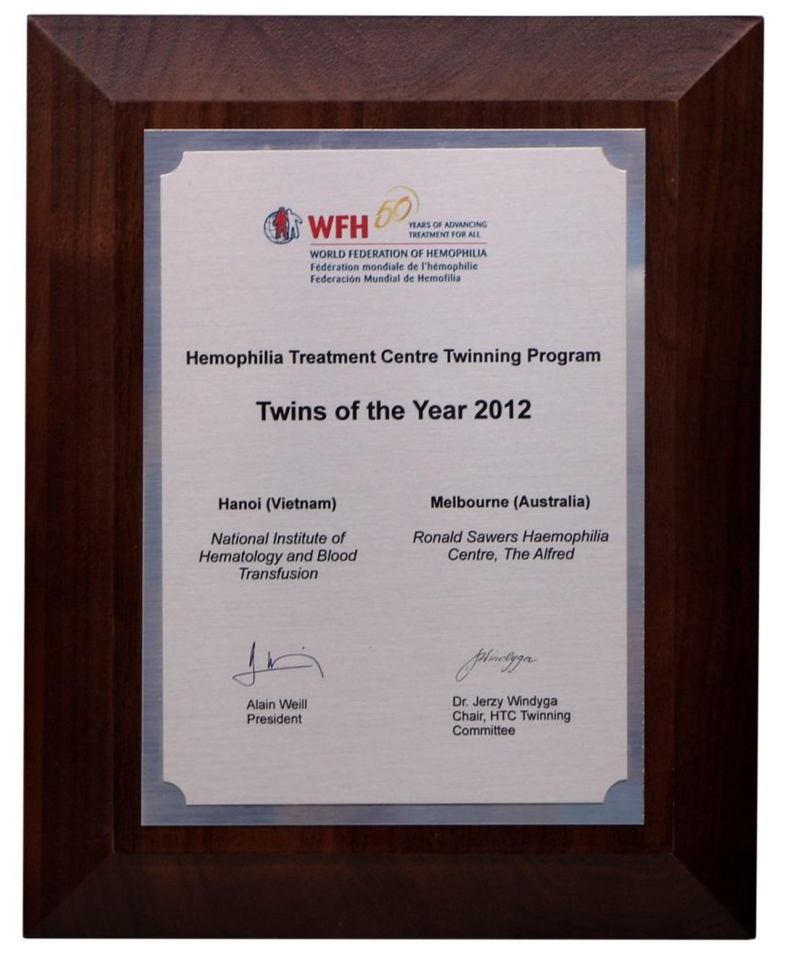
WFH
Hemophilia Treatment Centre Twinning Program
Twins of the Year 2012
Hanoi (Vietnam)
Melbourne (Australia)

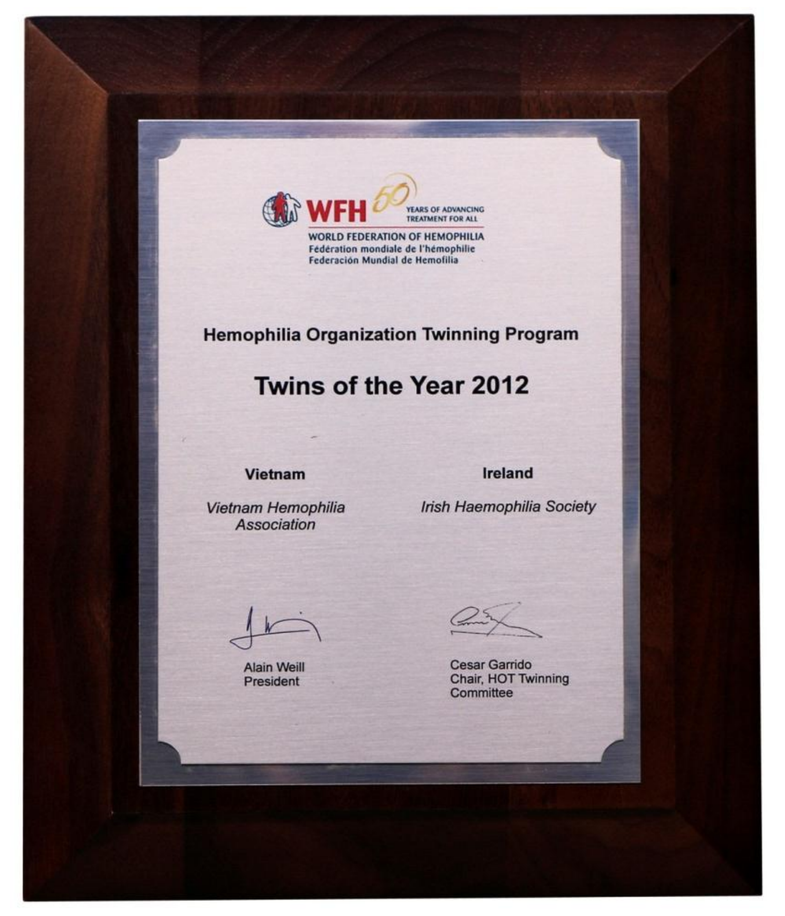
WFH
Hemophilia Organization Twinning Program
Twins of the Year 2012
Vietnam
Ireland

CONCLUSION:

In the future, the strategy of VHA is: Reorganization hemophilia care system, integrated into the health system; Training and education; Enhancing blood products in quantity, quality and variety; Promote the activities of patient associations; International cooperation; advocacy.

PosterSessionOnline.com
Poster Presented at: WFH 2016 World Congress
DOI: 10.3252/pso.eu.WFH2016.2016
Hemophilia Programs and Organizations
Mai Nguyen
48--PP-M
WFH2016