MOLECULAR BASIS OF FACTOR VII DEFICIENCY IN A CANADIAN POPULATION STUDY

GAUTHIER J[1], SMITH VC[2], PARSYAK S[2], DE SANTIS B[2], LU A[2], CURTIS SB[2], COUTURE F[1], NAVA T[2], WU JK[2], RIVARD GE[1] AND MACGILLIVRAY RTA[2]

[1]CHU Sainte-Justine, Molecular Diagnostic Laboratory, Université de Montréal, [2]Centre for Blood Research, University of British Columbia

TOPIC: RARE BLEEDING DISORDERS

INTRODUCTION

Factor VII (FVII) deficiency is an inherited autosomal recessive bleeding disorder caused by diminution or absence of blood coagulation FVII. FVII levels are regulated by environmental and genetic factors. Both the genetic basis and the biological pattern are highly heterogeneous. FVII deficiency is identified by a plasma FVII activity below 50%. Molecular analysis of the *F7* gene confirms the diagnosis but its interpretation is sometimes challenging. Over 250 unique mutations in the *F7* gene have been described. Some polymorphisms, mainly in the promoter region, influence the FVII plasma level thereby complicating the interpretation of molecular findings. Amongst these polymorphisms, six (Figure 1) are of physiological relevance and in part account for FVII plasma level variations. Herein we present preliminary results on *F7* gene sequencing and FVII plasma levels in a cohort of subjects that are currently attending the CHU Sainte-Justine haemostasis clinic. Bleeding scores have been obtained for some of them.

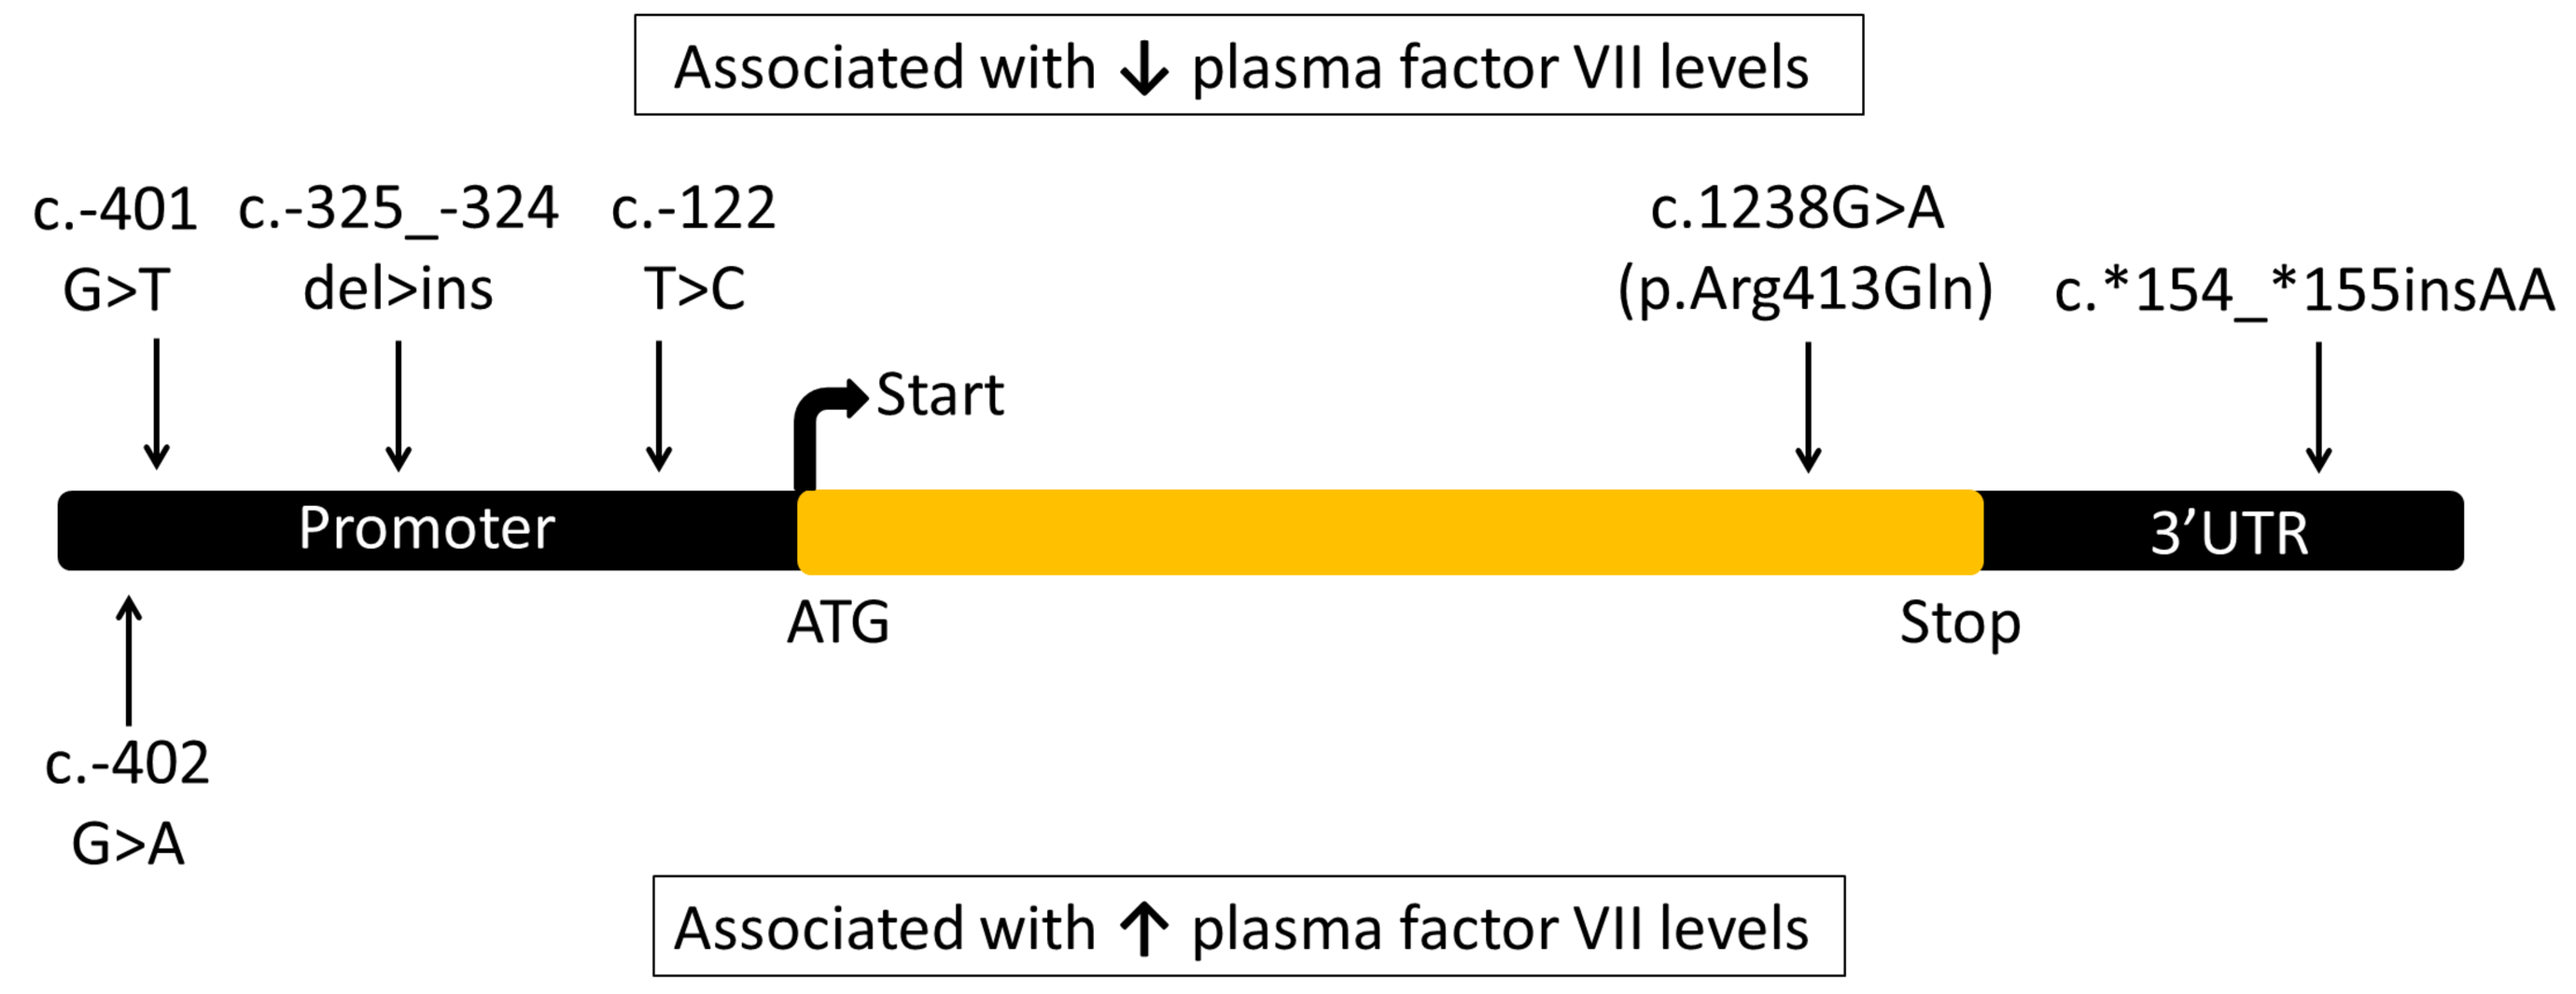
Associated with ↓ plasma factor VII levels
c.-401 G>T
c.-325_-324 del>ins
c.-122 T>C
c.1238G>A (p.Arg413Gln)
c.*154_*155insAA
Start
Promoter
3'UTR
ATG
Stop
c.-402 G>A
Associated with ↑ plasma factor VII levels

Figure 1. Schematic representation of *F7* gene and the genomic locations of the six genetic variants reported as modulating FVII plasma levels (ref. 1-5).

OBJECTIVES

To investigate the molecular basis of FVII deficiency in subjects and their relatives.

MATERIALS & METHODS

Our cohort includes 76 DNA samples; 45 of them are from probands referred for investigation of FVII deficiency. We determined the *F7* gene sequences from at least two family members in 19 families and DNA of 26 singletons with FVII deficiency. The *F7* gene sequences including splicing sites, promoter region (425 bp 5'upstream of exon 1) and the 3'UTR (210 bp) were determined for each subject and results were analyzed with the Mutation Surveyor software package. Large deletion/duplication analysis was ruled out using the digital droplet PCR system. FVII dosage was performed using the one-stage clotting assay using rabbit brain thromboplastin. Bleeding tendency was assessed with the ISTH Bleeding Assessment Tool (ref.6).

RESULTS

A total of 76 distinct DNA samples were analysed for the entire *F7* gene. Plasma FVII activity levels were available for 72/76 of them including 15 patients with <50% FVII activity and 11 individuals with a FVII level between 50-70%. In addition to the six functional and benign variants, we detected fifteen different genetic variants known to be associated with factor VII deficiency (Figure 2). At the present time, bleeding scores have been obtained for 15/76 patients.

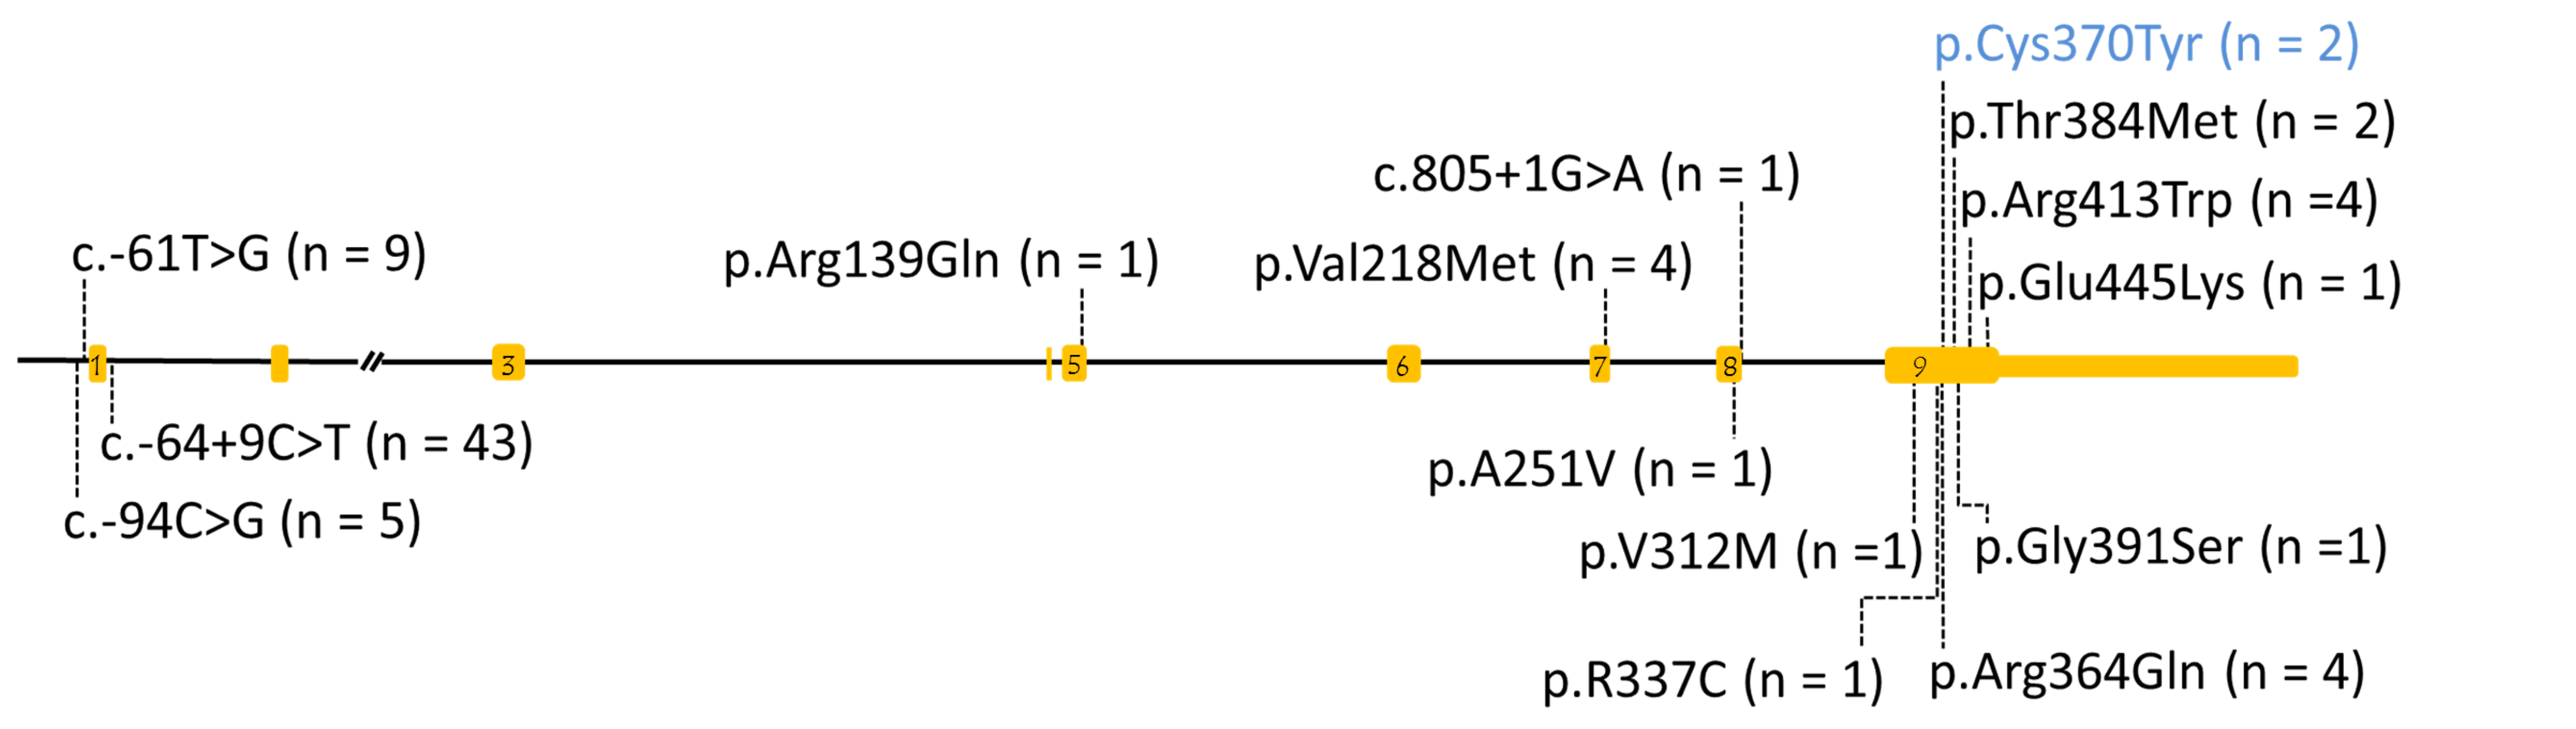
p.Cys370Tyr (n = 2)
p.Thr384Met (n = 2)
p.Arg413Trp (n =4)
p.Glu445Lys (n = 1)
c.805+1G>A (n = 1)
c.-61T>G (n = 9)
p.Arg139Gln (n = 1)
p.Val218Met (n = 4)
1
3
5
6
7
8
9
c.-64+9C>T (n = 43)
c.-94C>G (n = 5)
p.A251V (n = 1)
p.V312M (n =1)
p.Gly391Ser (n =1)
p.R337C (n = 1)
p.Arg364Gln (n = 4)

Figure 2. Genetic variants identified in 76 subjects. The variants c.-94C >G and c.-61T>G are located in the promoter region. In blue, p.Cys370Tyr, is to our knowledge a newly described variant. The number of subjects harboring each variant is indicate in parenthesis.

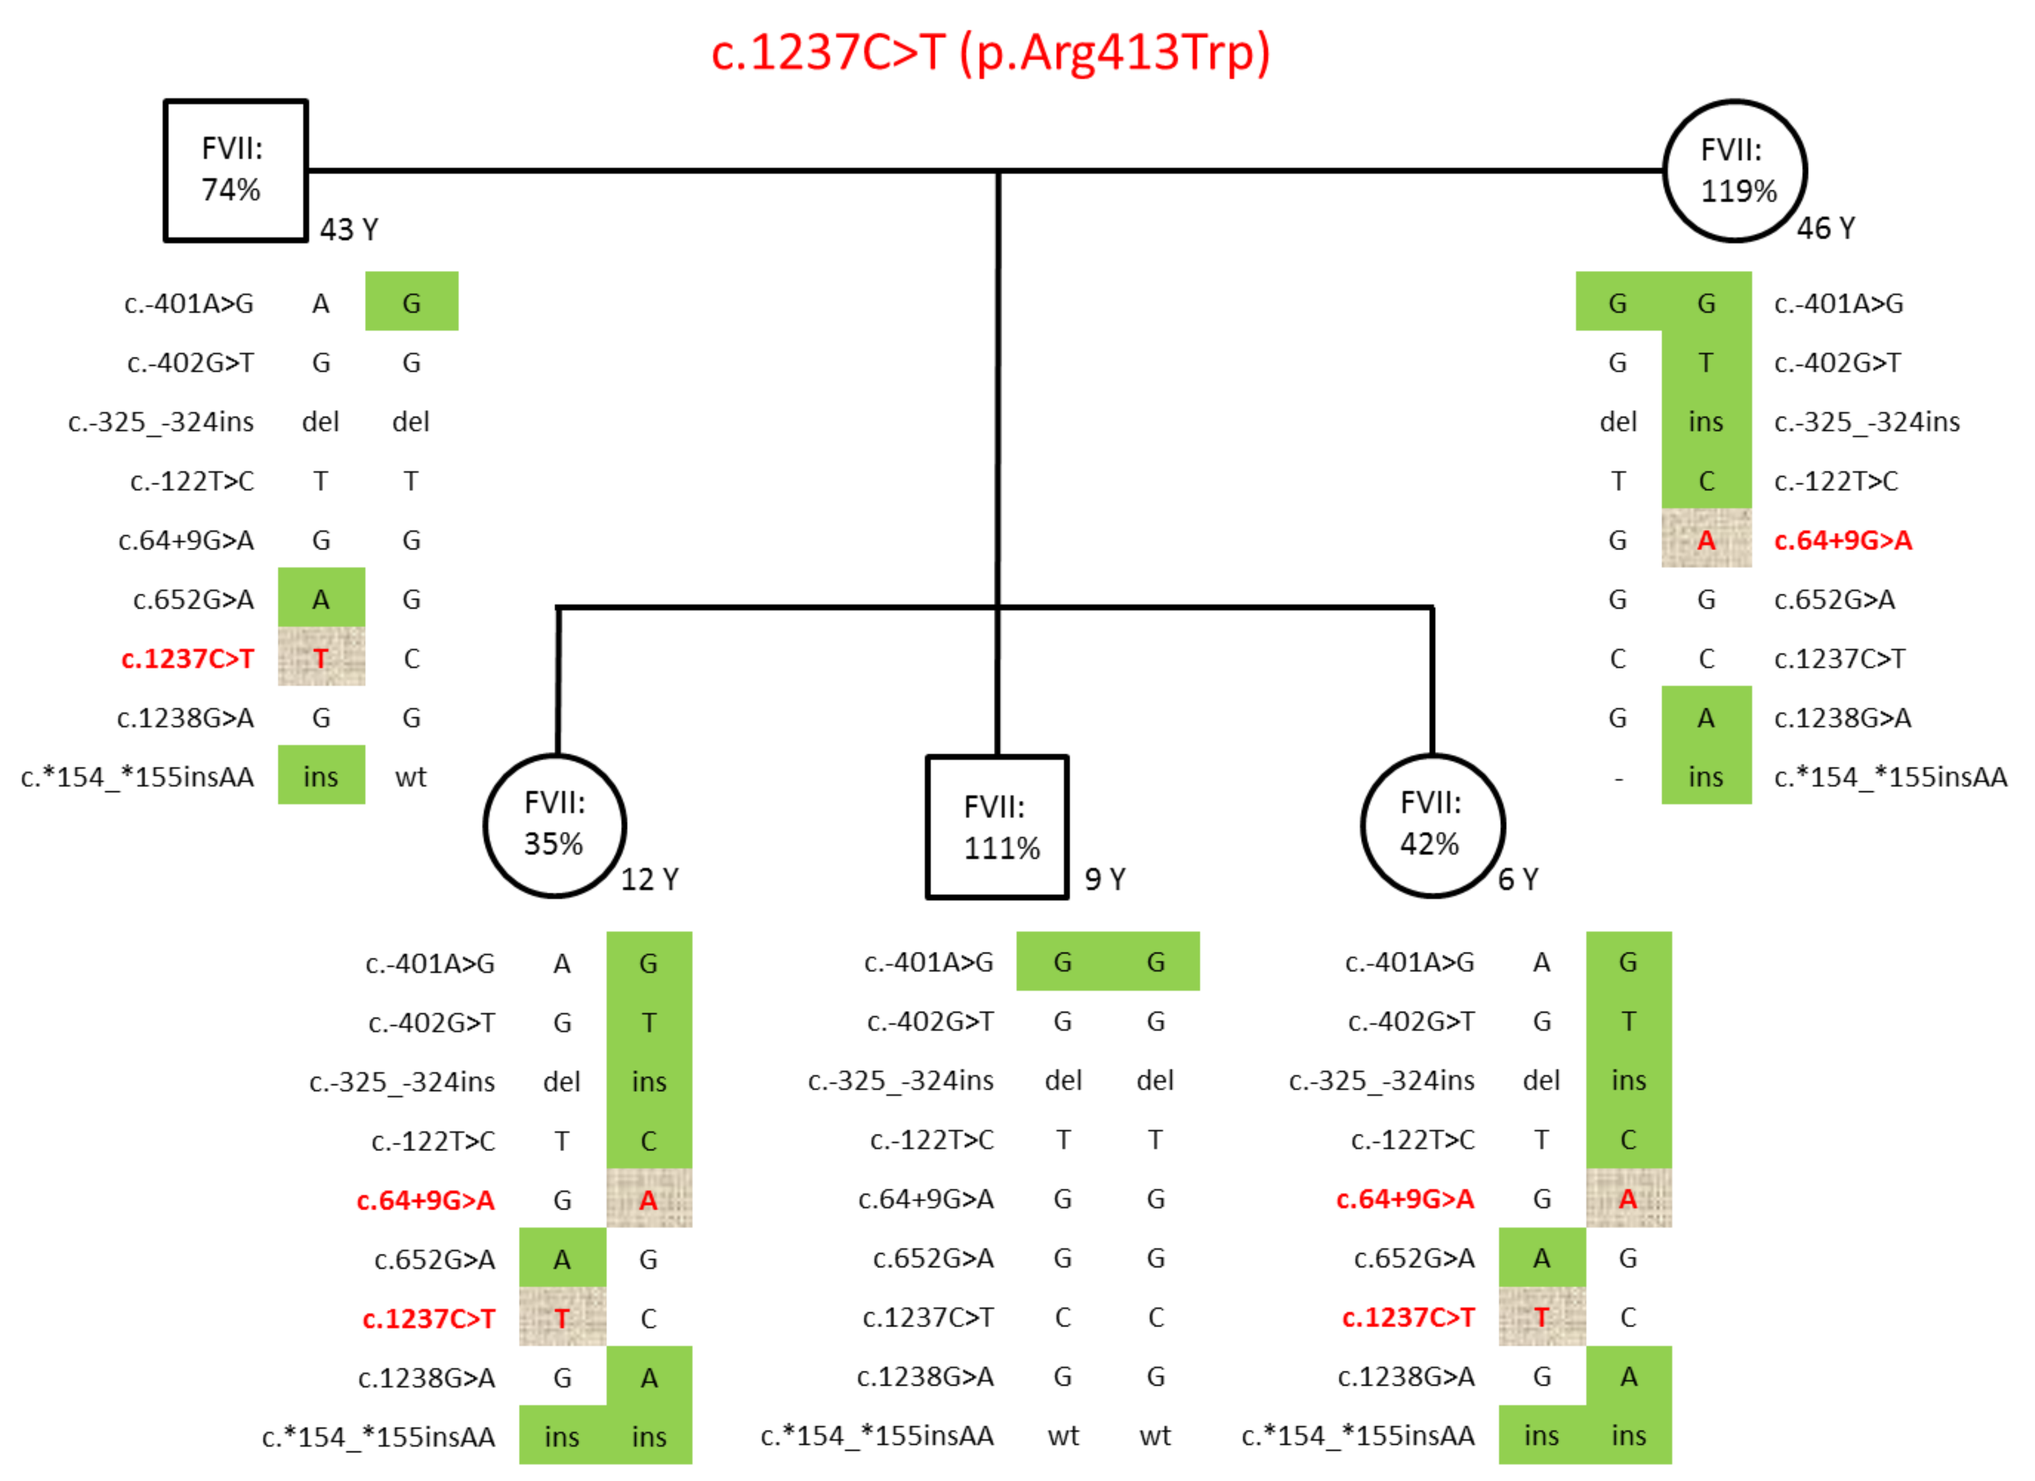
c.1237C>T (p.Arg413Trp)
FVII: 74%
43 Y
FVII: 119%
46 Y
FVII: 35%
12 Y
FVII: 111%
9 Y
FVII: 42%
6 Y

Figure 3. Pedigree of a family with the **c.1237C>T** (p.Arg413Trp) variant. Both the father and his daughters are heterozygous for **c.1237C>T** but have significantly different FVII plasma levels (74% vs 35% and 42%). The contributing maternal allele, harboring the **six functional polymorphisms** plus the **c.64+9 G>A** variant probably play a role in the observed lower FVII plasma levels in the daughters.

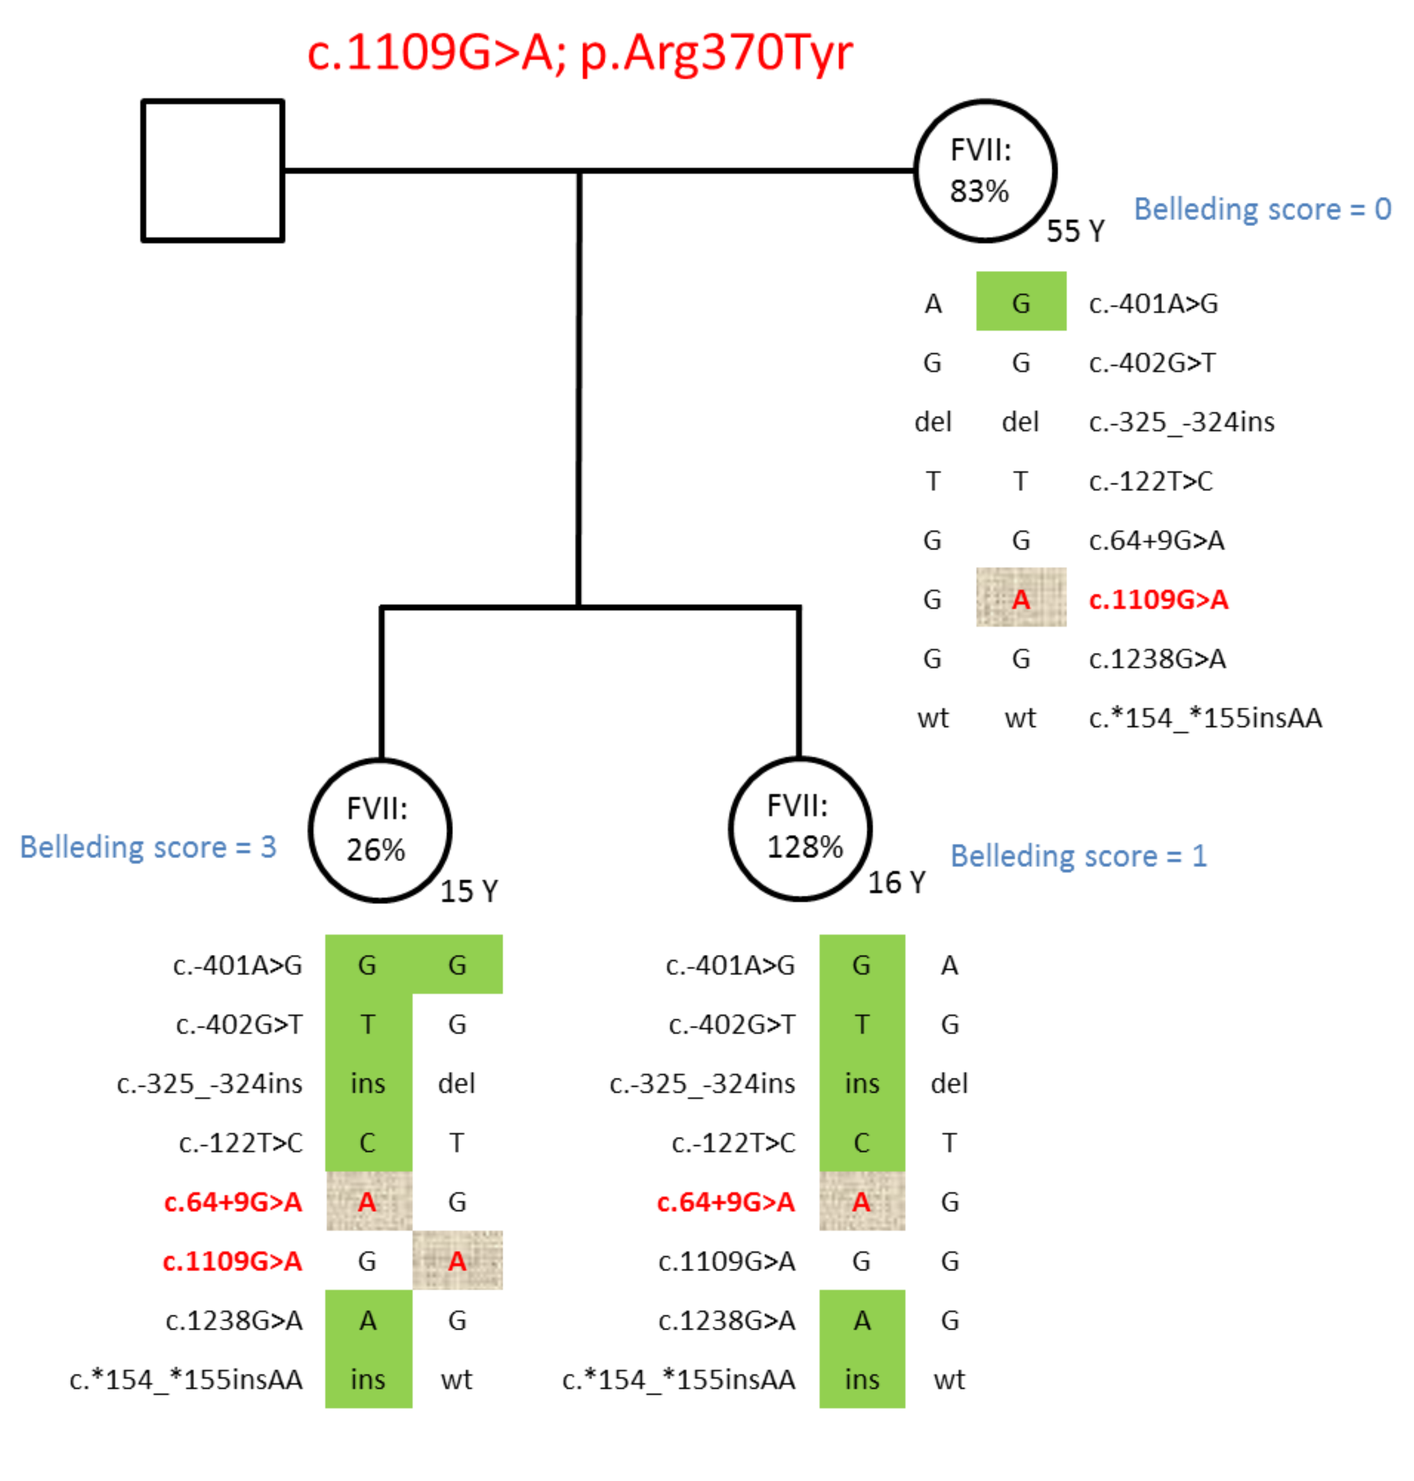
c.1109G>A; p.Arg370Tyr
FVII: 83%
55 Y
Belleding score = 0
FVII: 26%
15 Y
Belleding score = 3
FVII: 128%
16 Y
Belleding score = 1

Figure 4. Pedigree of a family with the novel **c.1237C>T** (p.Arg370Tyr) variant. This variant is transmitted from the asymptomatic mother (FVII 83%) to her symptomatic (FVII 26%) daughter. On the parental allele, the daughter inherited all **six functional polymorphisms** known to decrease FVII plasma level.

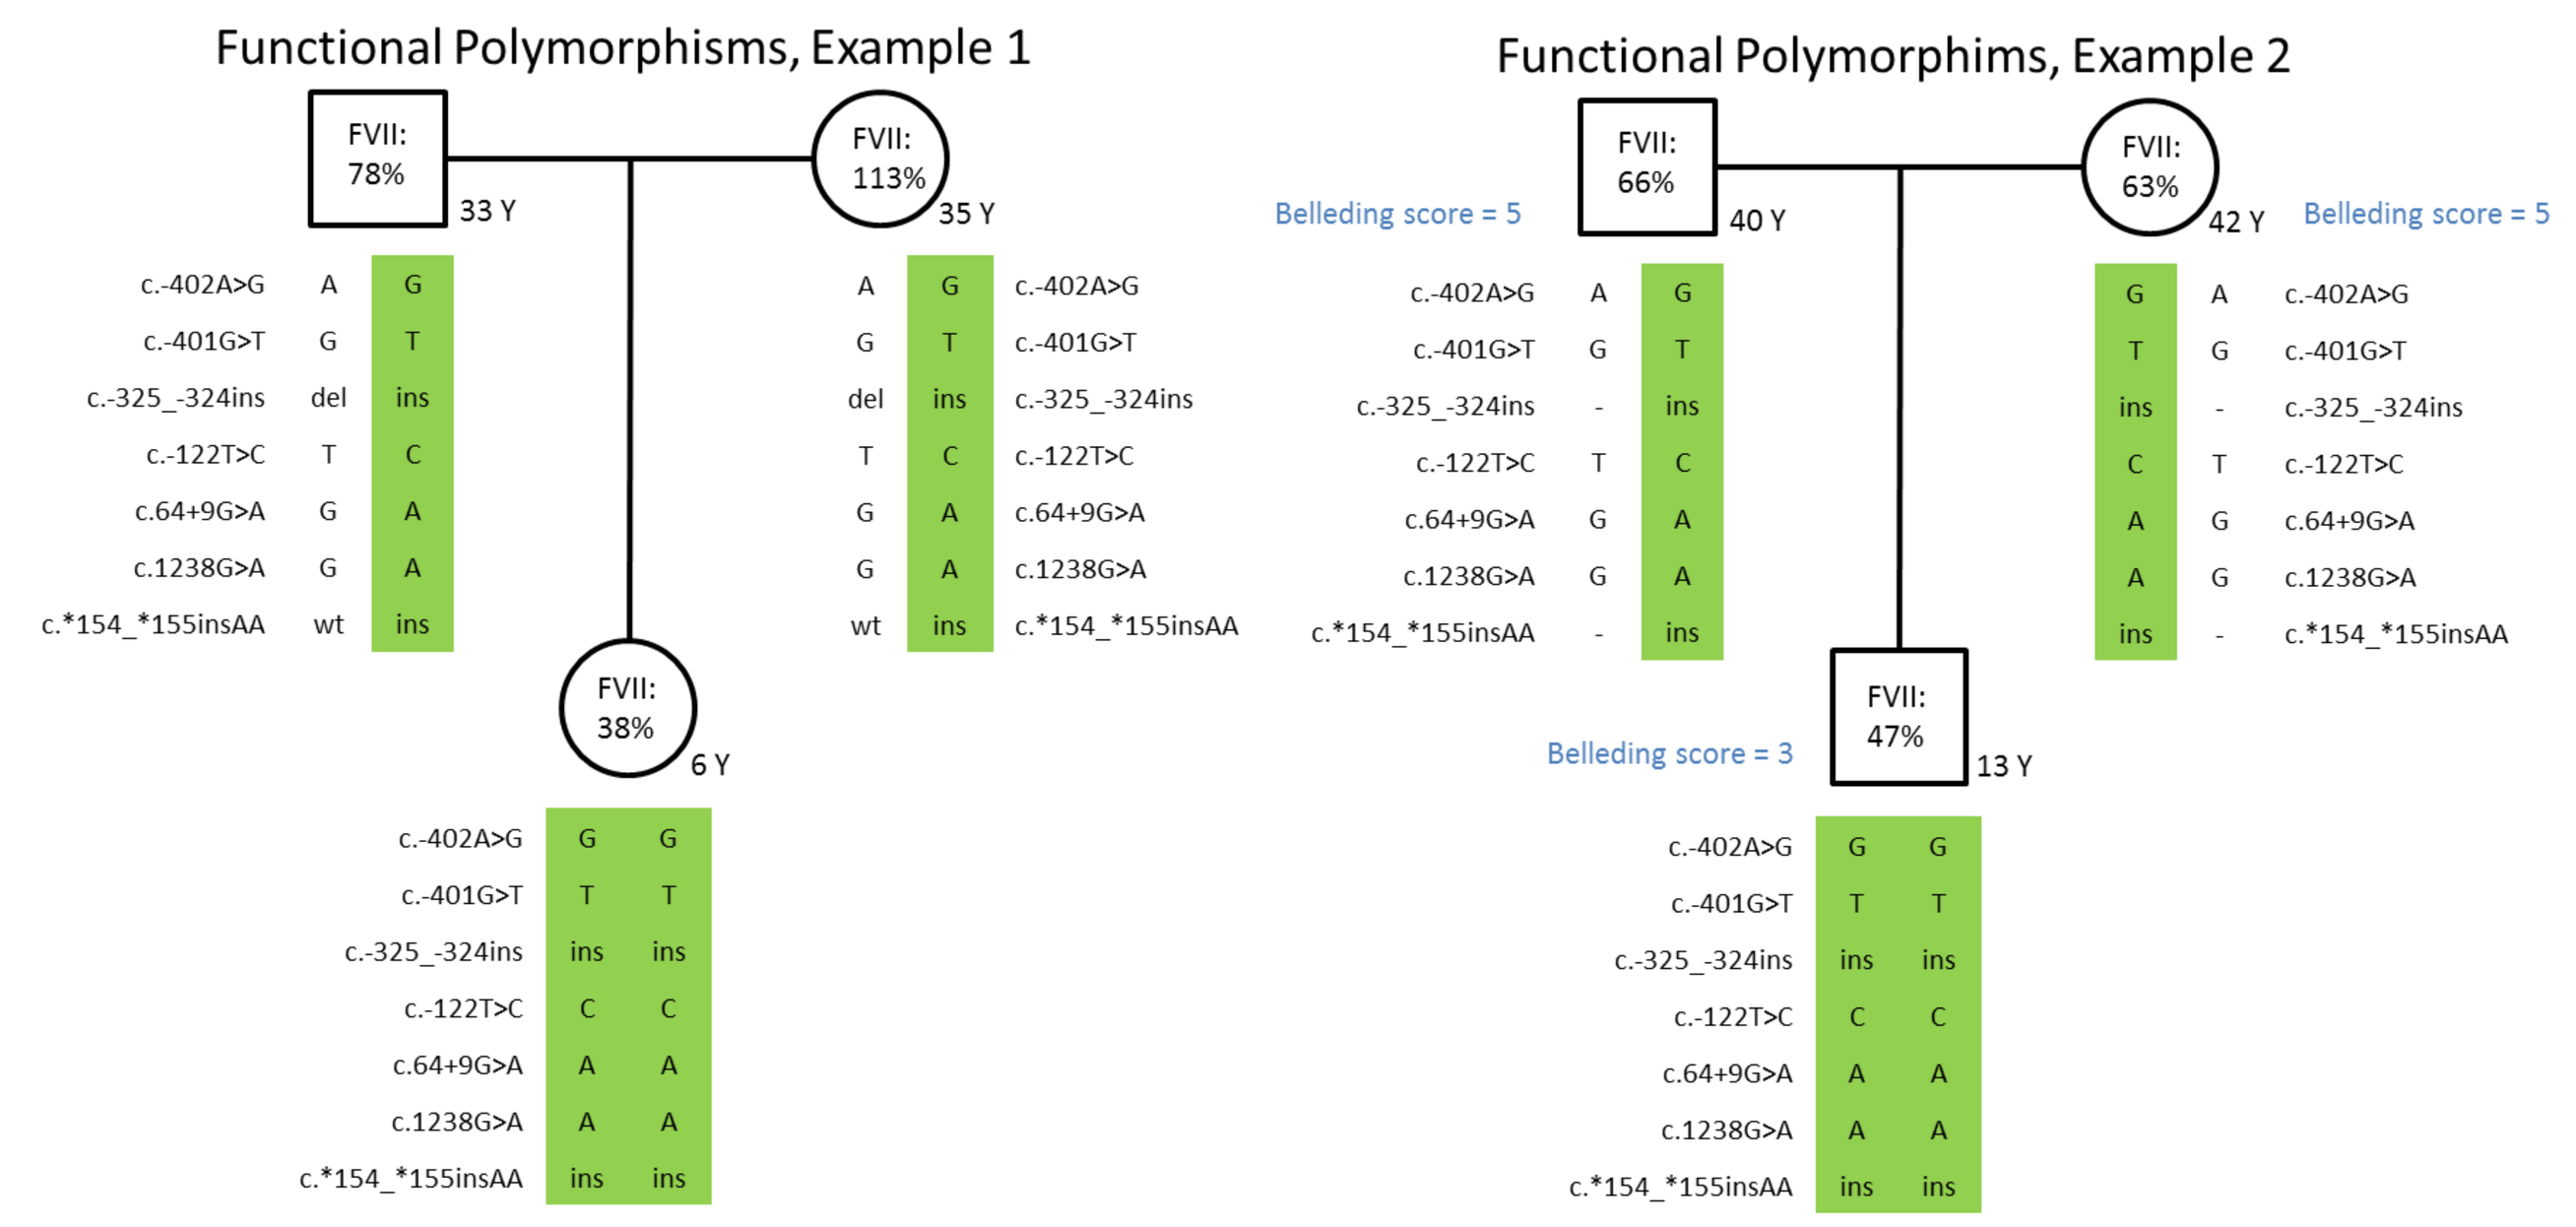
Functional Polymorphisms, Example 1
FVII: 78%
33 Y
FVII: 113%
35 Y
FVII: 38%
6 Y
Functional Polymorphims, Example 2
FVII: 66%
40 Y
Belleding score = 5
FVII: 63%
42 Y
Belleding score = 5
FVII: 47%
13 Y
Belleding score = 3

Figure 5. Examples of pedigrees demonstrating the effect of polymorphisms in a homozygous state. Both homozygous patients are negative for large deletions or duplications of the *F7* gene.

DISCUSSION

We sequenced the entire coding regions and boundaries of the *F7* genes in 45 unrelated subjects with FVII deficiency and their relatives. Fifteen genetic variants were identified in addition to the known functional polymorphisms. All identified variants had already been reported except for c.1109G>A (p.Arg370Tyr). For all but one subject with FVII deficiency, the low plasma FVII level could be explain by a *F7* mutation in combination with the functional polymorphisms. One subject (FVII of 37%) appeared to be « normal » at the gene screening level. After further investigation, a large deletion including part of the *F7* and *F10* genes was detected by CGH array analysis (data not shown). Our preliminary observations indicate that FVII plasma levels are sometimes highly influenced by the six functional polymorphisms as exemplified by the pedigrees presented in Figure 5. Other variants that are known to modulate factor VII levels such as -670 A>C and -630 A>G in the promoter region and IVS7 need to be investigated for a more complete assessment of the genetic contribution to FVII plasma levels in this cohort.

CONCLUSION

For FVII deficient subjects, the search for a causative mutation(s) in the *F7* gene is not sufficient to make a molecular diagnosis. Intra-genic and extra-genic functional polymorphisms of *F7* should be included in the molecular investigation.

REFERENCES

1. van't Hoof et al 1999. 2. Pollack et al. 1996. 3. Bernardi et al, 1996. 4. Hunautl et al 1997. 5. Peyvandi et al 2005. 6. ISTH adapted from de Rodeghiero, F. et al. ISTH/SSC Bleeding Assessment Tool: a standardized questionnaire and a proposal for a new bleeding score for inherited bleeding disorders. Journal of Thrombosis and Haemostasis, 8: 2063–2065 (Last Revision: 19 July 2011).

Supported by Bayer Canada

Poster Presented at:
DOI: 10.3252/pso.eu.WFH2016.2016
Rare bleeding disorders
Clemence Merlen
205--PP-M
WFH2016