HAEMOcare: an international epidemiological study of musculoskeletal disease burden in haemophilia patients in developing countries

Johnny Mahlangu,[1] Naresh Gupta,[2] Soraya Benchikh El Fegoun,[3] Meriem Belhani,[4] Khadija Maani,[5] Hossam Ali Saad,[6] Yasser Wali[7]

[1]Haemophilia Comprehensive Care Centre, Charlotte Maxeke Johannesburg Hospital, and Department of Molecular Medicine and Haematology, National Health Laboratory Service and Faculty of the Health Sciences, University of Witwatersrand, Johannesburg, South Africa; [2]Haemophilia Centre, and Department of Medicine, Maulana Azad Medical College and Associated Lok Nayak and GB Pant Hospitals, New Delhi, India; [3]Novo Nordisk Health Care AG, Zurich, Switzerland; [4]Department of Haematology and Blood Banking, University Hospital of Beni Messous, Algeria; [5]Department of Paediatrics, Haemophilia Centre, Haematology and Oncology, Abderrahim Harouchi Children's Hospital of Casablanca, Casablanca, Morocco; [6]Novo Nordisk Canada Inc., Toronto, Canada; [7]Department of Child Health, Sultan Qaboos University, Muscat, Oman

Introduction

- Reducing musculoskeletal disease burden remains an unmet healthcare need in haemophilia.[1]
- Patients with inhibitors generally report greater morbidity than those without.[2]
- In developed countries with access to treatment and prophylaxis, studies have evaluated orthopaedic status and outcomes such as quality of life (QoL) and resource consumption to improve disease management in patients without inhibitors.[2–7]
- With prophylaxis and optimal treatment of haemophilia, musculoskeletal disease burden and QoL are better when compared with developing countries.
- The primary objective of the HAEMOcare study was to evaluate orthopaedic status in severe haemophilia patients in developing countries.
- In the five countries where the study was conducted, access to prophylaxis and immune tolerance induction (ITI) was not available for all patients. More than 90% of patients, with or without inhibitors, were not using prophylactic therapy; only one young patient with inhibitors was receiving ongoing low-dose ITI.

Materials and methods

Study design

- HAEMOcare (ClinicalTrials.gov: NCT01503567) was a non-interventional, cross-sectional, multicentre, observational study conducted in developing countries (Algeria, India, Morocco, Oman and South Africa).[8]
- The study was approved by local institutional review boards/independent ethics committees and approval obtained accordingly.
- Written informed consent was obtained from each patient or their legally acceptable representative, before enrolment.
- Every patient had one visit during which all study related assessments were conducted; the end of the observational study was defined as the completion (last visit) of the last patient.

Patients

- Eligible patients were males aged ≥6 years with severe congenital haemophilia A or B (FVIII or FIX, respectively, <1 IU/dL or <1% of normal) with or without inhibitors, and treated on-demand for haemophilia.
- Individuals with other clinically relevant coagulation disorders, and those receiving treatment for hepatitis C or human immunodeficiency virus infection, were excluded.

Endpoints

- Orthopaedic status was evaluated using the Haemophilia Joint Health Score (HJHS) and Pettersson's score (higher scores represent worse status for both scales of measurement).
- QoL was assessed using the EuroQol 5-dimension questionnaire (EQ-5D-3L) and its Visual Analogue Scale (VAS).
- Economic burden was calculated for the 12 months before the study visit: direct and indirect expenses, and capacity to cover expenses (i.e. insurance status and socio-economic status of the patient/ family).
- For secondary objectives, patients were divided into four groups: paediatric (aged 6–18 years) without inhibitors; paediatric with inhibitors; adults without inhibitors; adults with inhibitors.

An electronic version of the poster can be viewed by scanning the Quick Response (QR) code. The QR code is intended to provide scientific information for personal use only. The poster should not be altered and may not be reproduced without permission from the authors.

Results

Patient population

- The study was conducted between 2nd January and 3rd September 2012.
- Of 282 patients (Table 1), 80 (28%) were from India, 60 (21%) each from Algeria and Morocco, 53 (19%) from Oman, and 29 (10%) from South Africa.
- Fifty patients had haemophilia A and inhibitors; there were no inhibitor-positive patients with haemophilia B.
- Of the 232 patients without inhibitors, 200 (86%) had haemophilia A and 32 (14%) had haemophilia B.
- Of 36 patients with inhibitors and available family history, 11 (31%) had a family history of inhibitors, compared with 9/157 patients (6%) without inhibitors.

Table 1 Demographic characteristics of the study population and pre-defined subgroups

	Paediatric (6–18 years)		Adult (>18 years)		
	Without inhibitors (n=104)	With inhibitors (n=24)	Without inhibitors (n=128)	With inhibitors (n=26)	Total (n=282)
Age (years)					
Mean (SD)	11.8 (3.6)	12.3 (3.8)	28.5 (9.2)	27.0 (5.8)	20.8 (10.6)
Race, n (%)					
Caucasian	54 (52)	14 (58)	48 (38)	12 (46)	128 (45)
Black/African	0	0	14 (11)	6 (23)	20 (7)
Asian	31 (30)	4 (17)	41 (32)	6 (23)	82 (29)
Other	19 (18)	6 (25)	25 (20)	2 (8)	52 (18)
Family history of haemophilia, n (%)					
Yes	66 (63)	17 (71)	91 (71)	19 (73)	193 (68)
No	38 (37)	7 (29)	37 (29)	7 (27)	89 (32)
Family history of inhibitors	(n=66)	(n=17)	(n=91)	(n=19)	(n=193)
Yes	6 (9)	5 (29)	3 (3)	6 (32)	20 (10)
No	58 (88)	10 (59)	86 (95)	12 (63)	166 (86)
Unknown	2 (3)	2 (12)	2 (2)	1 (5)	7 (4)
Type of haemophilia					
A	96 (92)	24 (100)	104 (81)	26 (100)	250 (89)
B	8 (8)	0	24 (19)	0	32 (11)
Time since diagnosis (months)	(n=100)	(n=23)	(n=123)	(n=22)	(n=268)
Mean (SD)	121.3 (43.1)	138.2 (44.2)	300.2 (122.1)	287.6 (97.8)	218.5 (126.4)
Time since diagnosis of inhibitors (months)		(n=24)		(n=25)	(n=49)
Mean (SD)	–	36.55 (27.45)	–	85.81 (68.37)	61.68 (57.59)
Average bleeds per month during prior 12 months	(n=86)	(n=22)	(n=124)	(n=26)	(n=258)
Mean (SD)	2.01 (1.77)	1.97 (1.66)	2.32 (2.03)	2.00 (1.51)	2.15 (1.87)
Presence of target joints, n (%)	89 (86)	18 (75)	112 (88)	17 (65)	236 (84)
Insurance/incapacity benefits, n (%)					
Fully reimbursed	37 (36)	15 (63)	71 (55)	16 (62)	139 (49)
Partially reimbursed	10 (10)	3 (13)	7 (5)	2 (78)	22 (8)
Not reimbursed	57 (55)	6 (25)	50 (39)	8 (31)	121 (43)

Table 2 Summary of global gait assessment (HJHS)

	Paediatric (6–18 years)		Adult (>18 years)		
	Without inhibitors (n=104)	With inhibitors (n=24)	Without inhibitors (n=128)	With inhibitors (n=26)	Total (n=282)
Mean (SD) HJHS global gait score	1.04 (1.19)	0.59 (1.10)	1.94 (1.29)	2.00 (1.26)	1.51 (1.33)
Skills not within normal limits, n (%)					
0	47 (45.2)	15 (62.5)	20 (15.6)	3 (11.5)	85 (30.1)
1	20 (19.2)	4 (16.7)	28 (21.9)	7 (26.9)	59 (20.9)
2	22 (21.2)	1 (4.2)	37 (28.9)	7 (26.9)	67 (23.8)
3	7 (6.7)	1 (4.2)	21 (16.4)	5 (19.2)	34 (12.1)
4	5 (4.8)	1 (4.2)	20 (15.6)	4 (15.4)	30 (10.6)
Not applicable/missing	3 (2.9)	2 (8.3)	2 (1.6)	0	7 (2.5)

Clinical assessments

- Overall, mean (standard deviation [SD]) HJHS was 17.86 (12.81) and observed Pettersson's score was 14.98 (13.47).
- Mean HJHS global gait score was 1.51 (1.33); approximately half of patients had all skills within normal limits or only one skill outside normal limits (Table 2).
- Overall, 236/281 patients (84%) had target joints, most commonly the knees (right 53%; left, 52%).
- No statistical difference in mean observed Pettersson score between inhibitor and non-inhibitor patients (adjusted estimated difference: 0.72 [95% confidence interval (CI): –3.91, 5.35]).
- No statistical difference in mean observed Pettersson score across subgroups (Figure 1).
- Mean (SD) observed Pettersson's score was 9.41 (1.80) in paediatric patients and 15.41 (1.81) in adults (adjusted estimated difference: 6.00 [95% CI: 2.47, 9.53]; p<0.001).

Figure 1 Mean observed Pettersson score among patients with and without inhibitors, and in the four pre-defined patient subgroups.

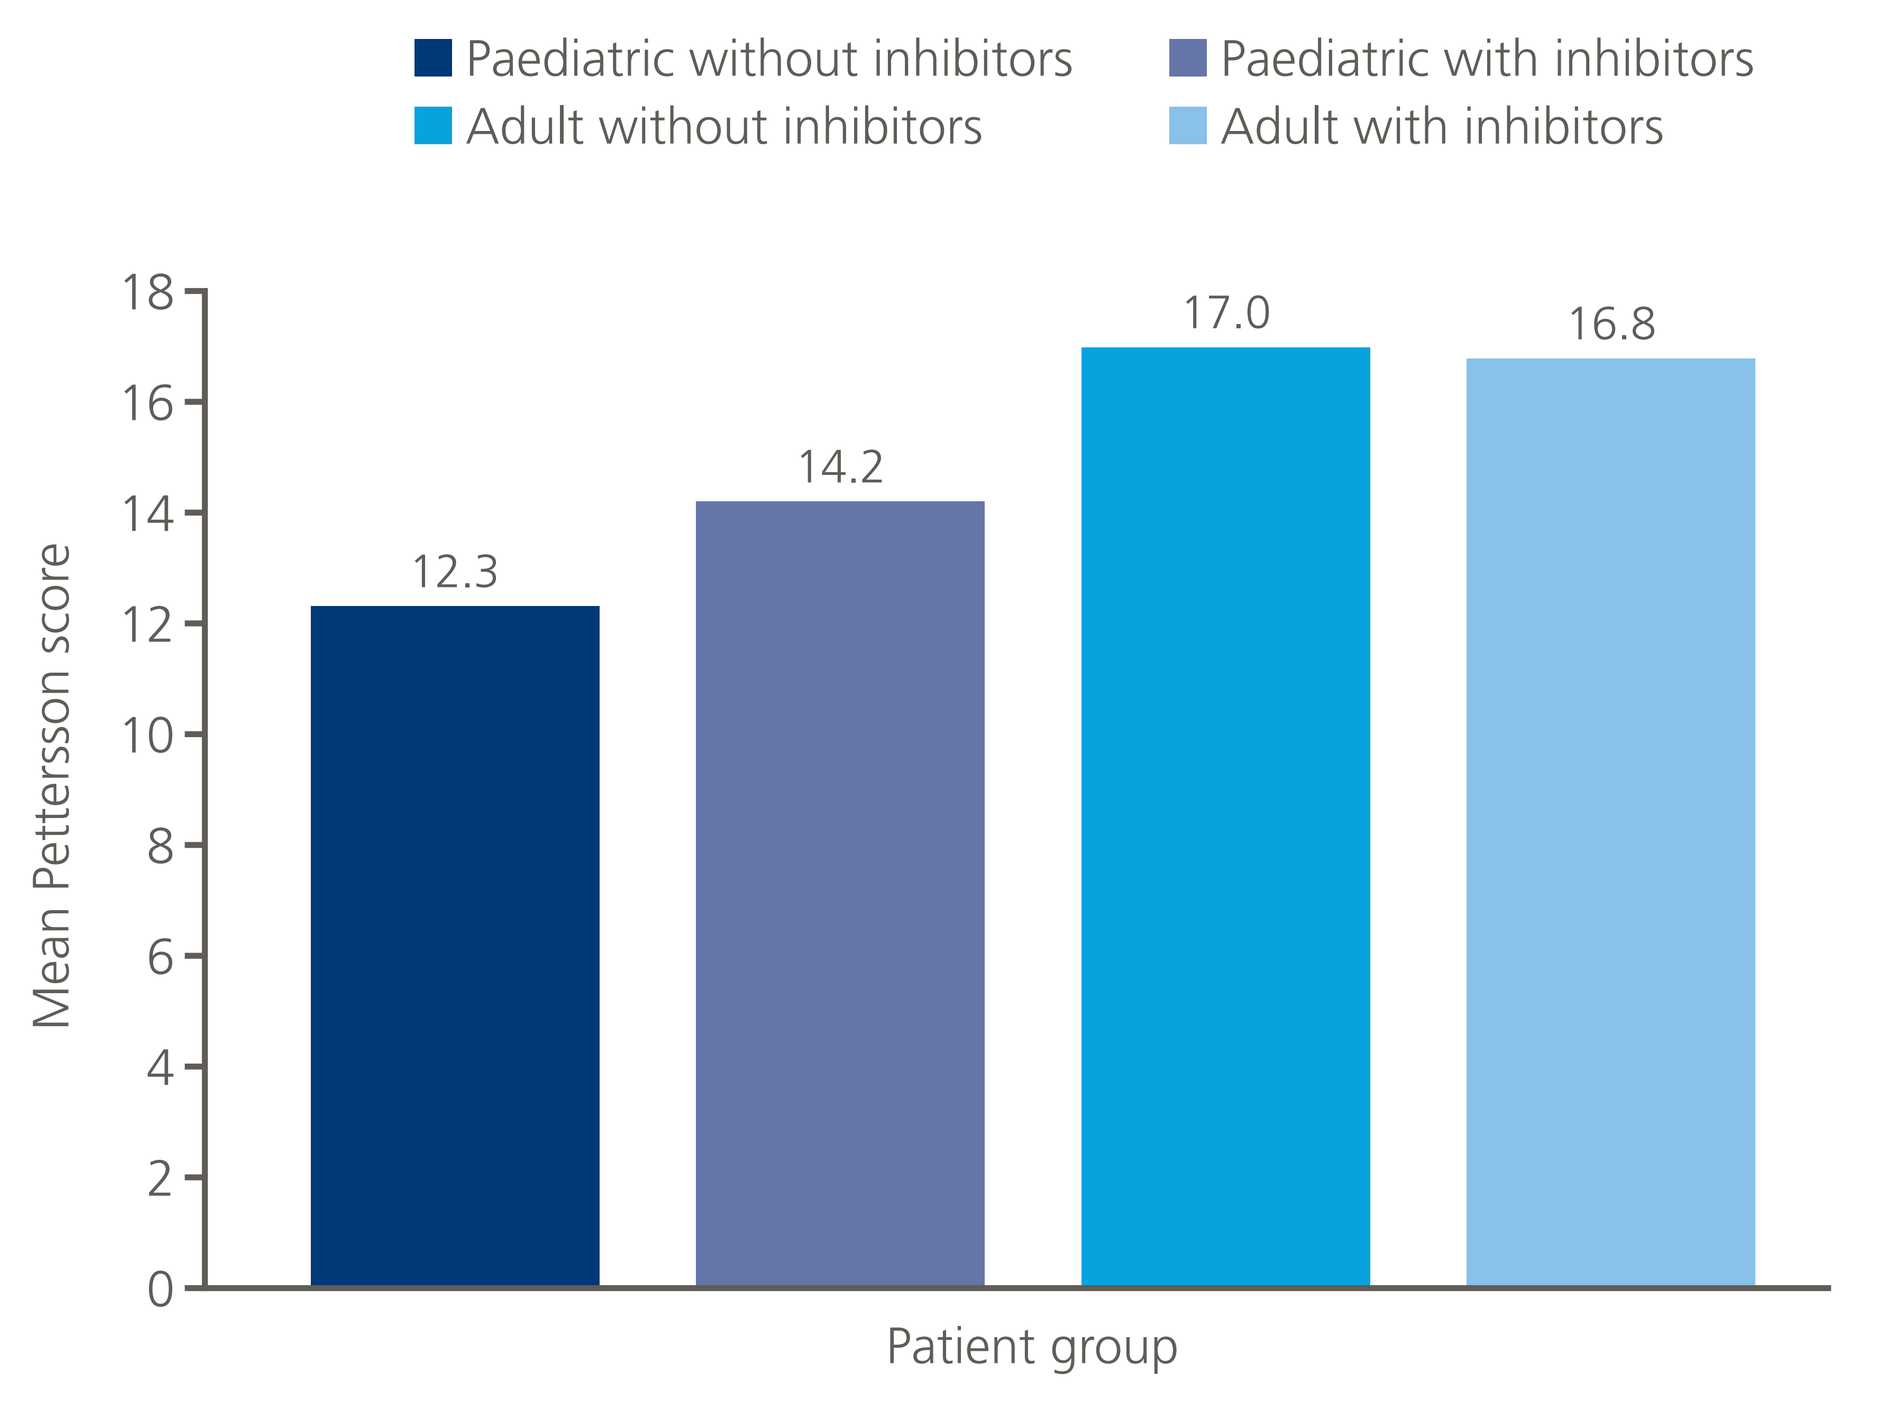
Paediatric without inhibitors
Paediatric with inhibitors
Adult without inhibitors
Adult with inhibitors
Mean Pettersson score
0
2
4
6
8
10
12
14
16
18
12.3
14.2
17.0
16.8
Patient group

- Mean HJHS was non-significantly higher in inhibitor versus non-inhibitor patients (adjusted estimated difference: –2.45 [95% CI: –6.30, 1.40]) (Figure 2).
- Mean (SD) HJHS was 10.19 (1.57) in paediatric patients and 17.25 (1.58) in adults (adjusted estimated difference: 7.05 [95% CI: 4.08–10.03]; p<0.001).

Figure 2 Mean total HJHS among patients with and without inhibitors, and in the four pre-defined patient subgroups.

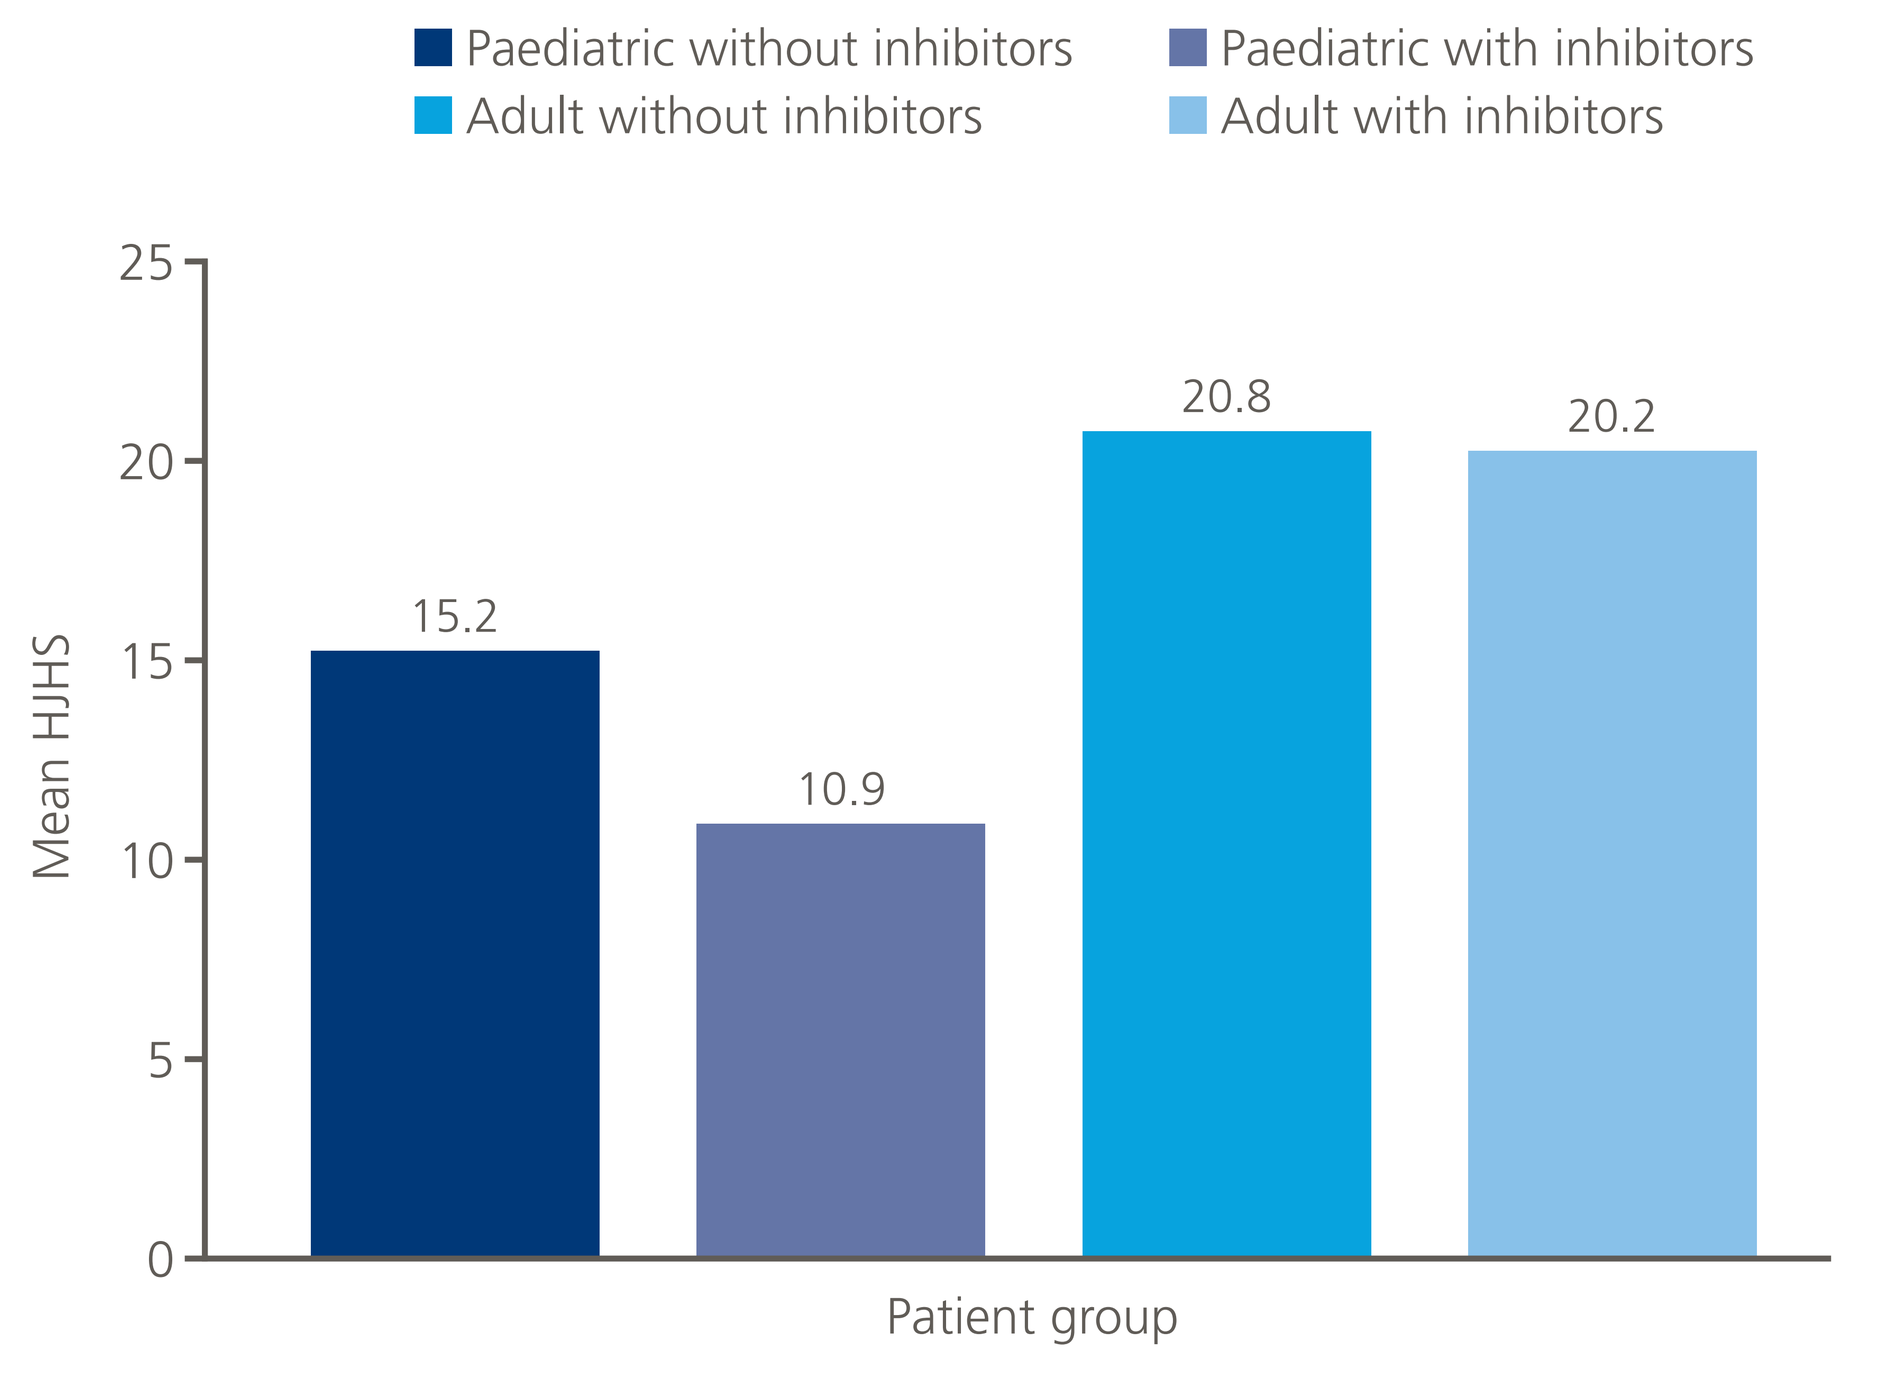
Paediatric without inhibitors
Paediatric with inhibitors
Adult without inhibitors
Adult with inhibitors
Mean HJHS
0
5
10
15
20
25
15.2
10.9
20.8
20.2
Patient group

Quality of life

- Many patients reported problems on the EQ-5D-3L, particularly in mobility and pain/discomfort; more adults than paediatric patients reported problems (Figure 3).
- Mean EQ-5D-3L VAS scores were better in patients without inhibitors (68.74) versus those with inhibitors (73.54), and ranged from 67.37 in adults without inhibitors to 76.67 in children with inhibitors.

Figure 3 Percentage of haemophilia patients who reported having problems according to the dimensions of the EQ-5D-3L questionnaire.

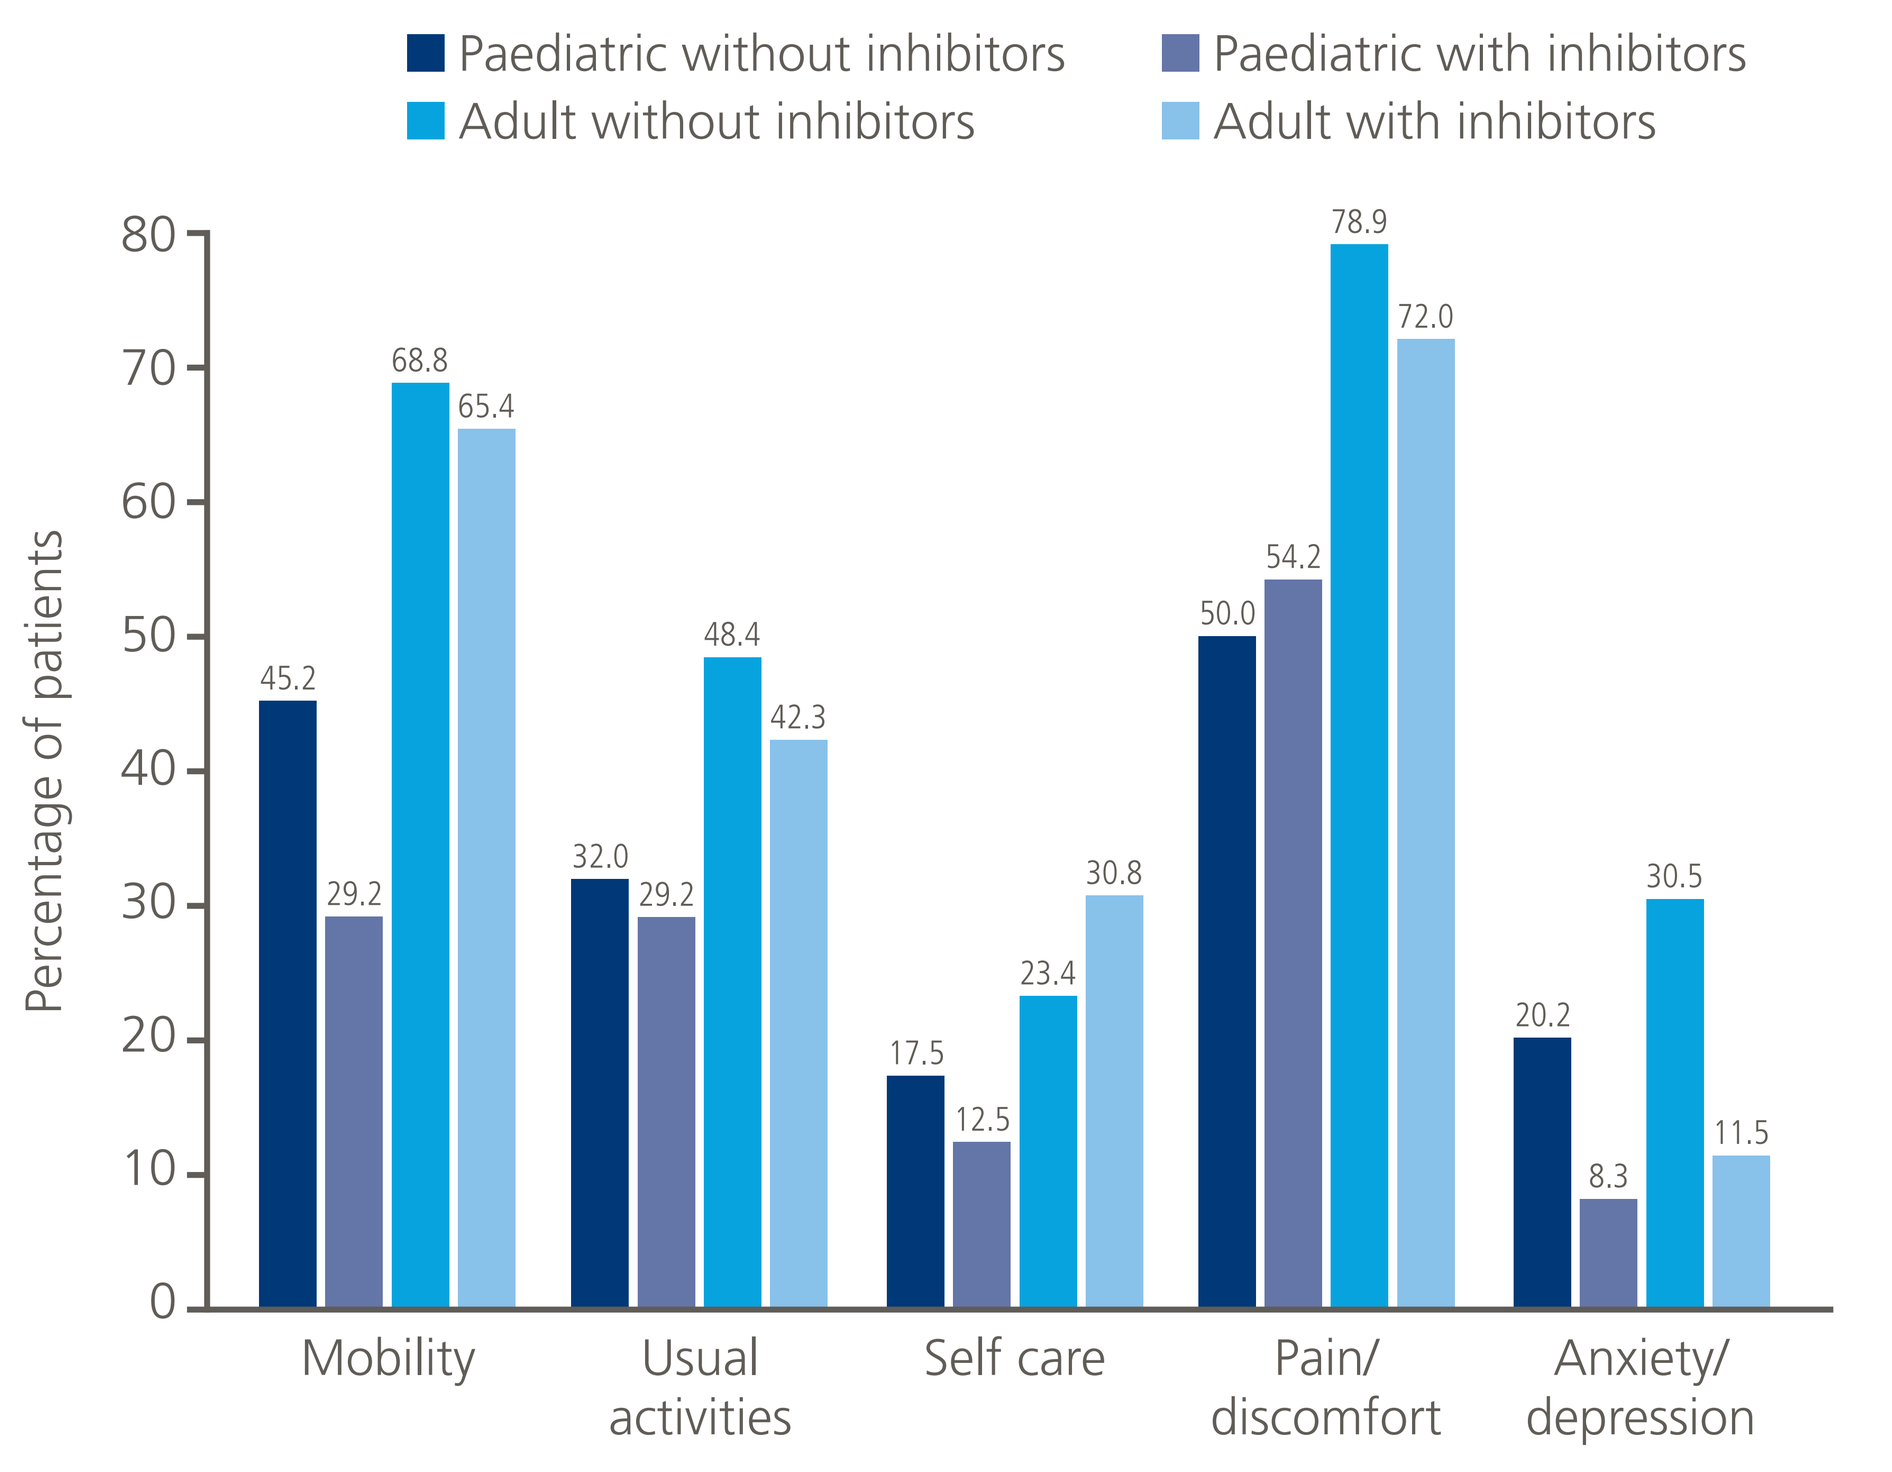
Paediatric without inhibitors
Paediatric with inhibitors
Adult without inhibitors
Adult with inhibitors
Percentage of patients
0
10
20
30
40
50
60
70
80
45.2
29.2
68.8
65.4
32.0
29.2
48.4
42.3
17.5
12.5
23.4
30.8
50.0
54.2
78.9
72.0
20.2
8.3
30.5
11.5
Mobility
Usual activities
Self care
Pain/ discomfort
Anxiety/ depression

Economic aspects of haemophilia

- Costs of treatment were fully reimbursed in 139/282 patients (49%), partially reimbursed in 22 patients (8%), and not reimbursed in 121 patients (43%).
- More patients with inhibitors were fully reimbursed (31/50; 62.0%) versus those without inhibitors (108/232; 47%).
- Median (range) family income/month was US$350 ($15–$7772).
- There were no significant differences in average monthly indirect costs between patients with and without inhibitors (Table 3).

Table 3 Mean (SD) indirect consumption of patient/family and community resources during the 12 months before enrolment.

	Without inhibitors	With inhibitors
	n=153	n=33
Days of school absenteeism (patient)	29.7 (36.7)	32.0 (36.1)
	n=42	n=8
Working days lost (patient)	30.8 (38.2)	69.5 (61.9)
Indirect cost of lost working days (US$)	440.9 (781.9)	3192.6 (4220.9)
	n=59	n=12
Working days lost (family)	19.7 (15.2)	25.8 (40.1)
Indirect cost of lost working days (US$)	344.0 (810.9)	415.6 (541.3)

Discussion

- In a study of similar design in 128 European males ≥14 years with haemophilia (with or without inhibitors), burden of orthopaedic complications and impact on QoL were more severe in patients with inhibitors than in those without.[2]
- Mean Pettersson scores were 27.8 in young adults (14–35 years) with inhibitors, 35.8 in older adults (36–65 years) with inhibitors, and 19.3 in younger adults without inhibitors;[2] these values are somewhat worse than reported in the present study (see Figure 1).

Conclusions

- **This is the first study documenting musculoskeletal burden of disease in developing countries. Despite the overall young age of the cohort, 84% had target joints.**
- **The design of HAEMOcare was inspired by a European study published 5 years before HAEMOcare was initiated.[2]**
- **The European study showed greater differences between patients with and without inhibitors than HAEMOcare, which showed very similar results between haemophilia with and without inhibitors. This highlights the need to improve haemophilia care in general.**
- **Improvements should include greater access to treatment, raised awareness of comprehensive care, and an enhanced role for the multidisciplinary team.**

References

1. Raffini L et al. *Br J Haematol* 2007;136:777–787.
2. Morfini M et al. *Haemophilia* 2007;13:606–612.
3. Tusell JM et al. *Haemophilia* 2002;8 (Suppl. 2):38–42.
4. Soucie JM et al. *Blood* 2004;103:2467–2473.
5. Windyga J et al. *Pol Arch Med Wewn* 2005;113:562–569.
6. Morfini M. *Haemophilia* 2008;14 (Suppl. 6):20–22.
7. Stieltjes N et al. *Blood Coagul Fibrinolysis* 2009;20:4–11.
8. Gupta N et al. Presented at the 18th Congress of the European Hematology Association, Stockholm, Sweden, June 13–16, 2013.

Disclosures

Johnny Mahlangu has received research grants from Bayer, Biogen, CLS, NovoNordisk and Roche. He is a member of advisory committees of Baxalta, Biogen, CLS, NovoNordisk and Roche, and speaker bureaux of Amgen, Bayer, Biogen, Biotest and CLS. Naresh Gupta, Meriem Belhani, Khadija Maani and Yasser Wali declare no conflict of interest. Soraya Benchikh El Fegoun is an employee of Novo Nordisk Health Care AG. Hossam Ali Saad is an employee of Novo Nordisk Canada Inc.

The HAEMOcare study was sponsored by Novo Nordisk. Medical writing support was provided by Dan Booth of Bioscript Medical Ltd and funded by Novo Nordisk.
Presented at the World Federation of Hemophilia World Congress, Orlando, FL, USA, July 24–28, 2016

PosterSessionOnline.com
Poster
Presented at:
WFH 2016 WORLD CONGRESS Orlando, USA July 24-28

DOI: 10.3252/pso.eu.WFH2016.2016
Orthopedic issues
Daniel Booth
121--PP-M
WFH2016