Poster 677

Long-Term Health Outcomes Comparison between Prophylaxis and On-Demand aPCC Treatment in Patients with Severe Haemophilia A with Factor VIII Inhibitors in Spain

Sabater E[1], Oyagüez I[1], Martin M[2], Bonanad S[3], Nuñez R[4], García JP[5], Casado MA[1]

[1] Pharmacoeconomics & Outcomes Research Iberia, Madrid, Spain; [2] Hospital Universitario La Paz, Madrid, Spain; [3] Hospital Universitario La Fe, Valencia, Spain; [4] Hospital Universitario Virgen del Rocío, Sevilla, Spain; [5] *Shire, Madrid, Spain.

INTRODUCTION

- The development of inhibitors complicates the management of hemophilia A due to the neutralizing effect of inhibitors on administered FVIII concentrates[1-2].
- Current treatment in patients with high-responding/high-titer inhibitors (≥ 5BU) or those with low-titer
- (< 5BU) inhibitors refractory to replacement therapy[3-5] requires the administration of bypassing agents such as FEIBA, an activated prothrombin-complex concentrate (aPCC)[6].
- The aPCC indications include on-demand treatment and prophylaxis of bleeds in haemophilia A patients with Factor VIII inhibitors.
- The available evidence[7-12] supports that the prophylactic use of aPCC, reduces the appearance of bleeding events compared with on-demand approach.
- The existing evidence is related to short-term follow-up periods. To cover longer periods modelling techniques, based on the available evidence, can be an alternative approach to estimate long-term outcomes.

OBJECTIVE

- To compare long-term outcomes between prophylaxis and on-demand regimens in patients with severe haemophilia A with FVIII Inhibitors treated with aPCC, through an extrapolation of the available evidence by means of modelling techniques.

MATERIALS AND METHODS

- A Markov model was developed to estimate health outcomes over a 40-year period. Cycle length was 1 week.
- Four health-states were established: alive with the presence of inhibitors, active bleeds (4 sub-states according bleeds location), surgery and death (Figure 1). Surgery state refers to potential surgical interventions required by disease complications
- The following health outcomes were estimated: number of bleeding episodes, life years gained (LYG) and quality-adjusted life years (QALYs) for any of the treatment strategies.
- Two treatment strategies, prophylaxis vs on-demand treatment of bleeding episodes, were compared.
- The model assessed two hypothetical cohorts of 100 HA adult patients treated with one of the strategies. The cohort mean age was 23.20 years[13].
- Proportions of bleeding episodes by location was obtained from literature[8].

Figure 1. Markov model structure

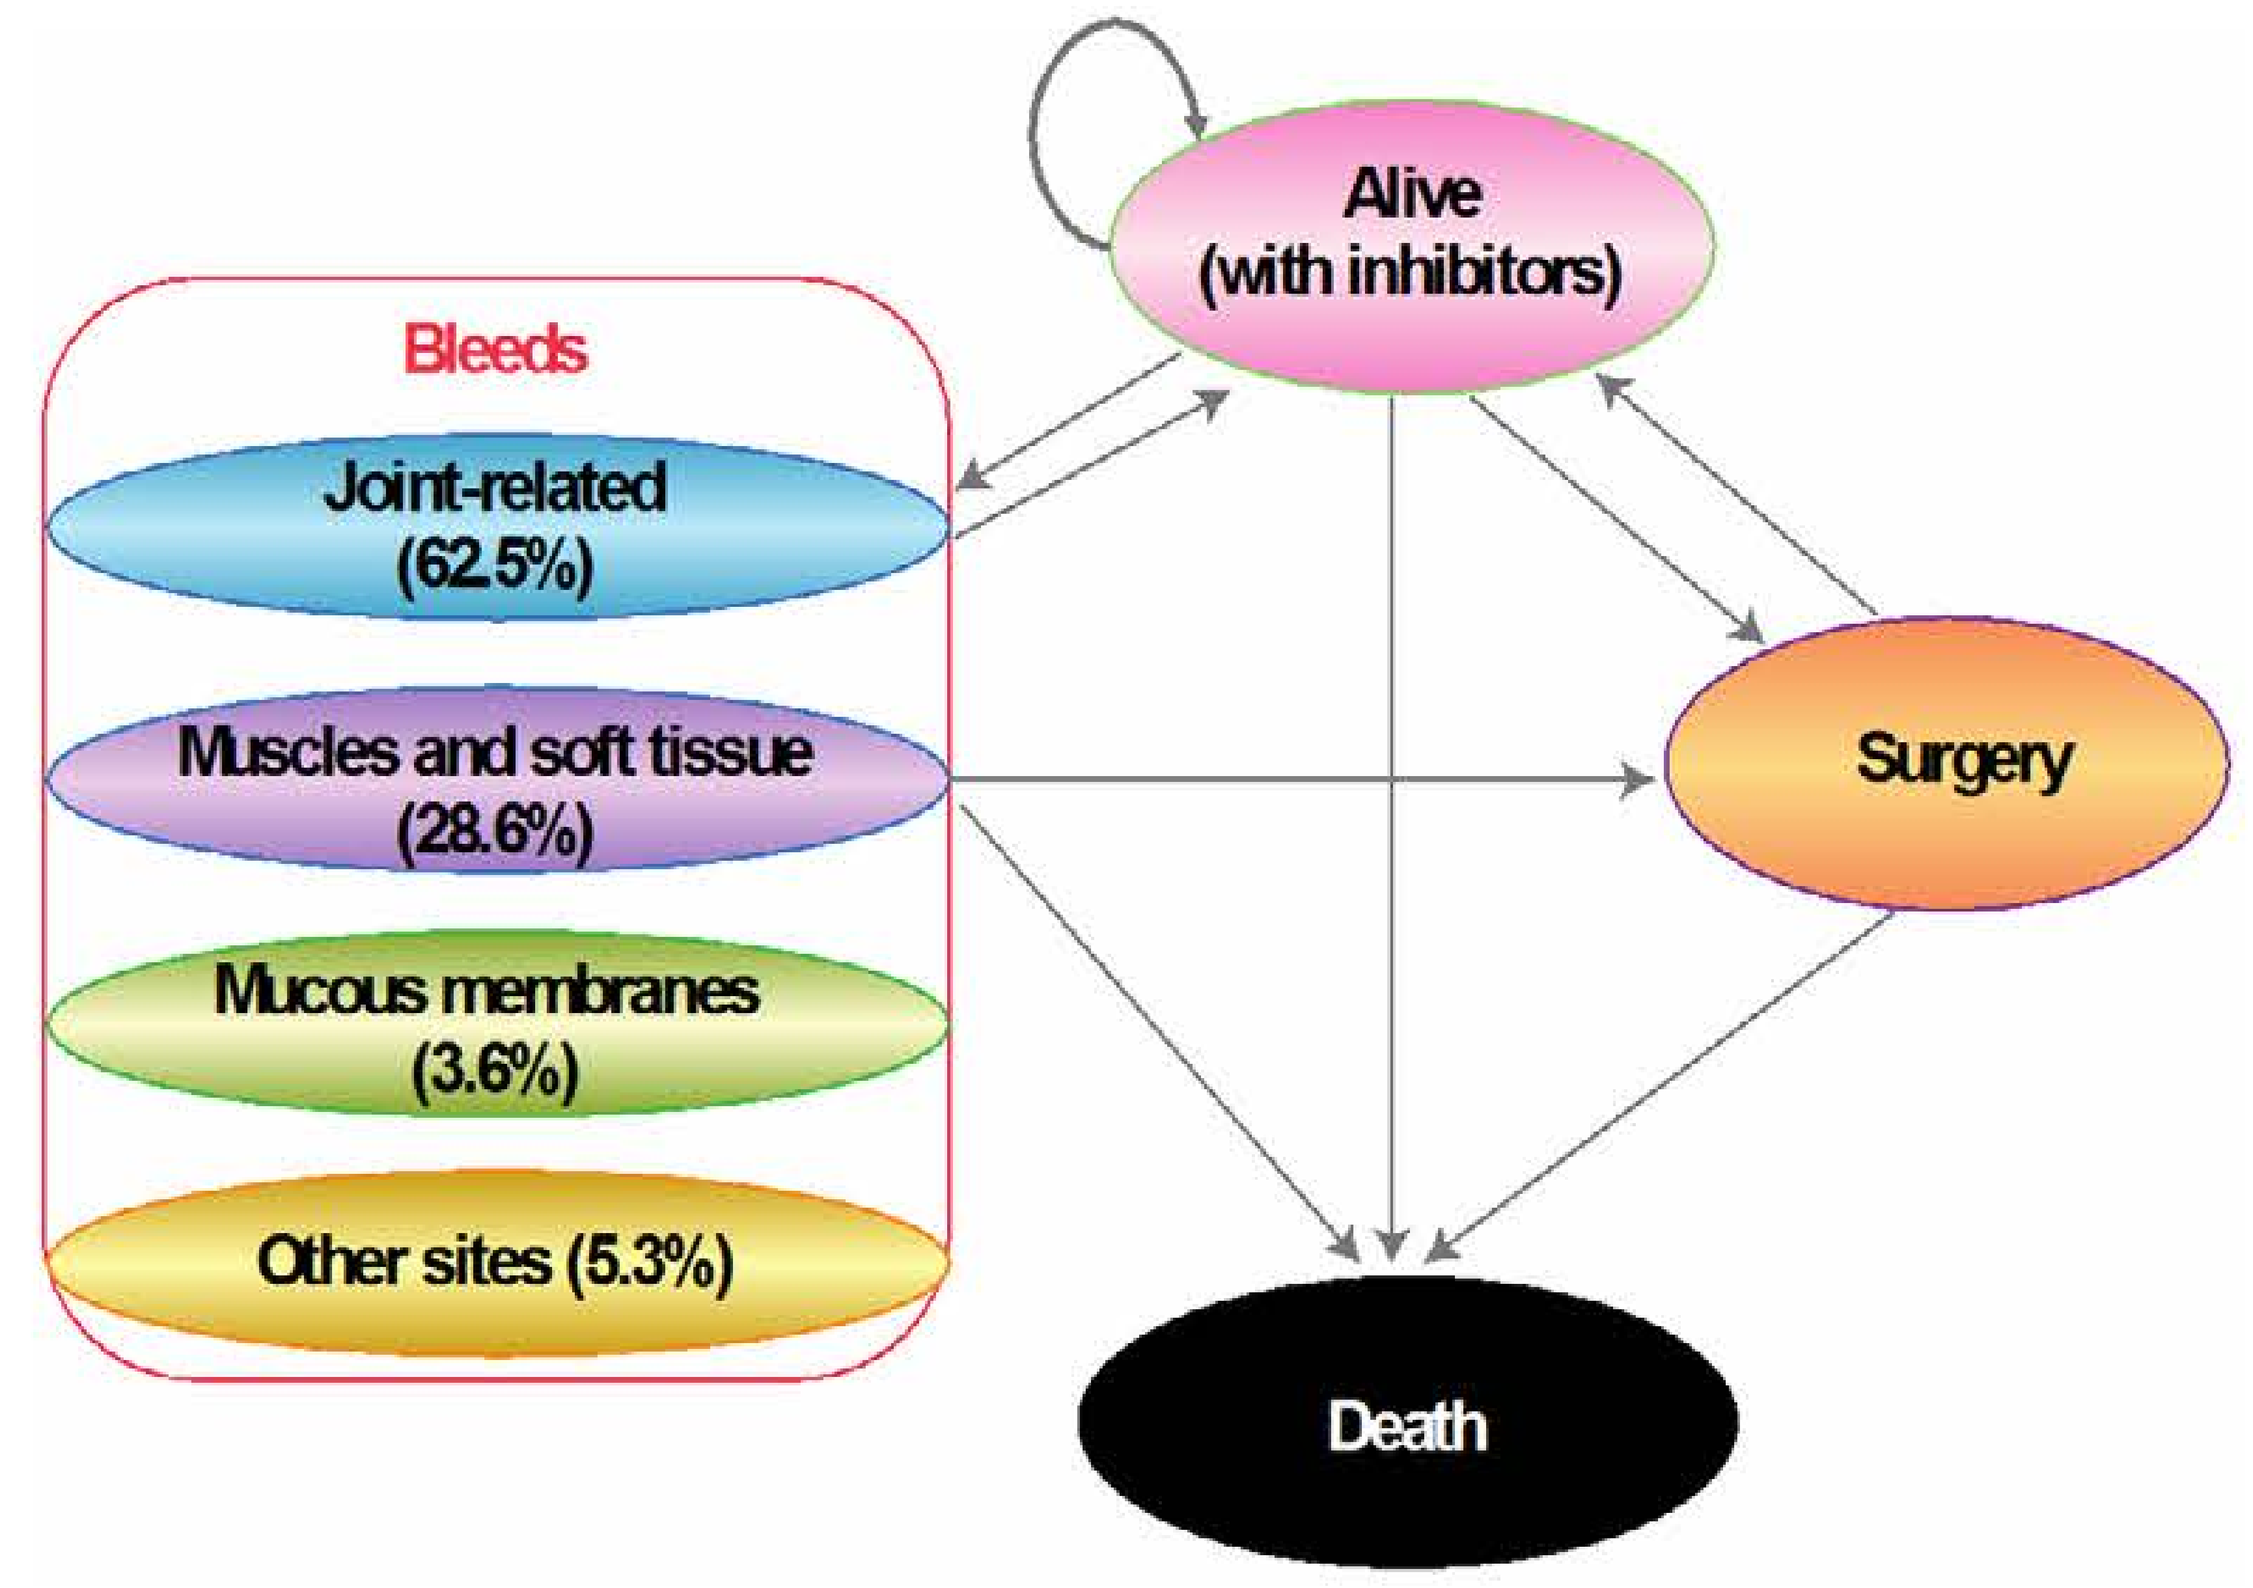
Alive
(with inhibitors)
Bleeds
Joint-related
(62.5%)
Muscles and soft tissue
(28.6%)
Mucous membranes
(3.6%)
Other sites (5.3%)
Surgery
Death

- Based on available evidence, prophylaxis strategy was associated to 68.65% reduction of annual frequency of bleeds[7-12], given an average of 8 bleeds per patient per year, of annual frequency of bleeds (25.23 per patient)[14].
- Annual rate of surgery (2.3%)[15] was assumed independent of bleed location or treatment strategy.
- Death probability derived from mortality age-standardized rates from Spanish National Statistics Institute[16]. An additional risk of death was applied to the surgery state, weekly increment was estimated based on annual rate of 0.023[15].
- Lineal models were applied to obtain the utility values for each health-state. The models and coefficients used were obtained from the literature[17-19].
- A probabilistic sensitivity analysis (PSA) was performed with 10,000 Monte-Carlo simulations. Model parameters varied using probability distributions (normal distributions for utility coefficients and for the average reduction of bleeds, Dirichlet distributions for bleed locations).
- PSA results were estimated as the median values of the simulations, with the correspondent 95% credible interval (95% CrI), for both avoided bleeds and incremental QALYS in patients treated with prophylaxis vs on-demand.

RESULTS

Base case results

- The base case analysis showed that at the end of the 40 years of simulation, the obtained mean number of bleeds for a patient treated with on-demand regimen was 983.
- Prophylaxis strategy avoided a mean of 673 bleeds per patient in comparison with on-demand treatment (Table 1). From these bleeds avoided, 62.5% were located in joints, 28.6% located in the muscles or soft tissue, 3.6% in mucous membranes and 5.3% in other locations (Figure 2).

Table 1. Base case analysis results

	On-Demand	Prophylaxis	Difference (propylaxis vs on-demand)
Bleeds	983 Bleeds	309 Bleeds	-673 Bleeds
Life years gained (LYG)	39.02 LYG	39.02 LYG	0.00 LYG
Quality-adjusted life years (QALY)	22.20 QALY	33.82 QALY	11.63 QALY

Figure 2. Location of avoided bleeds at 40 years

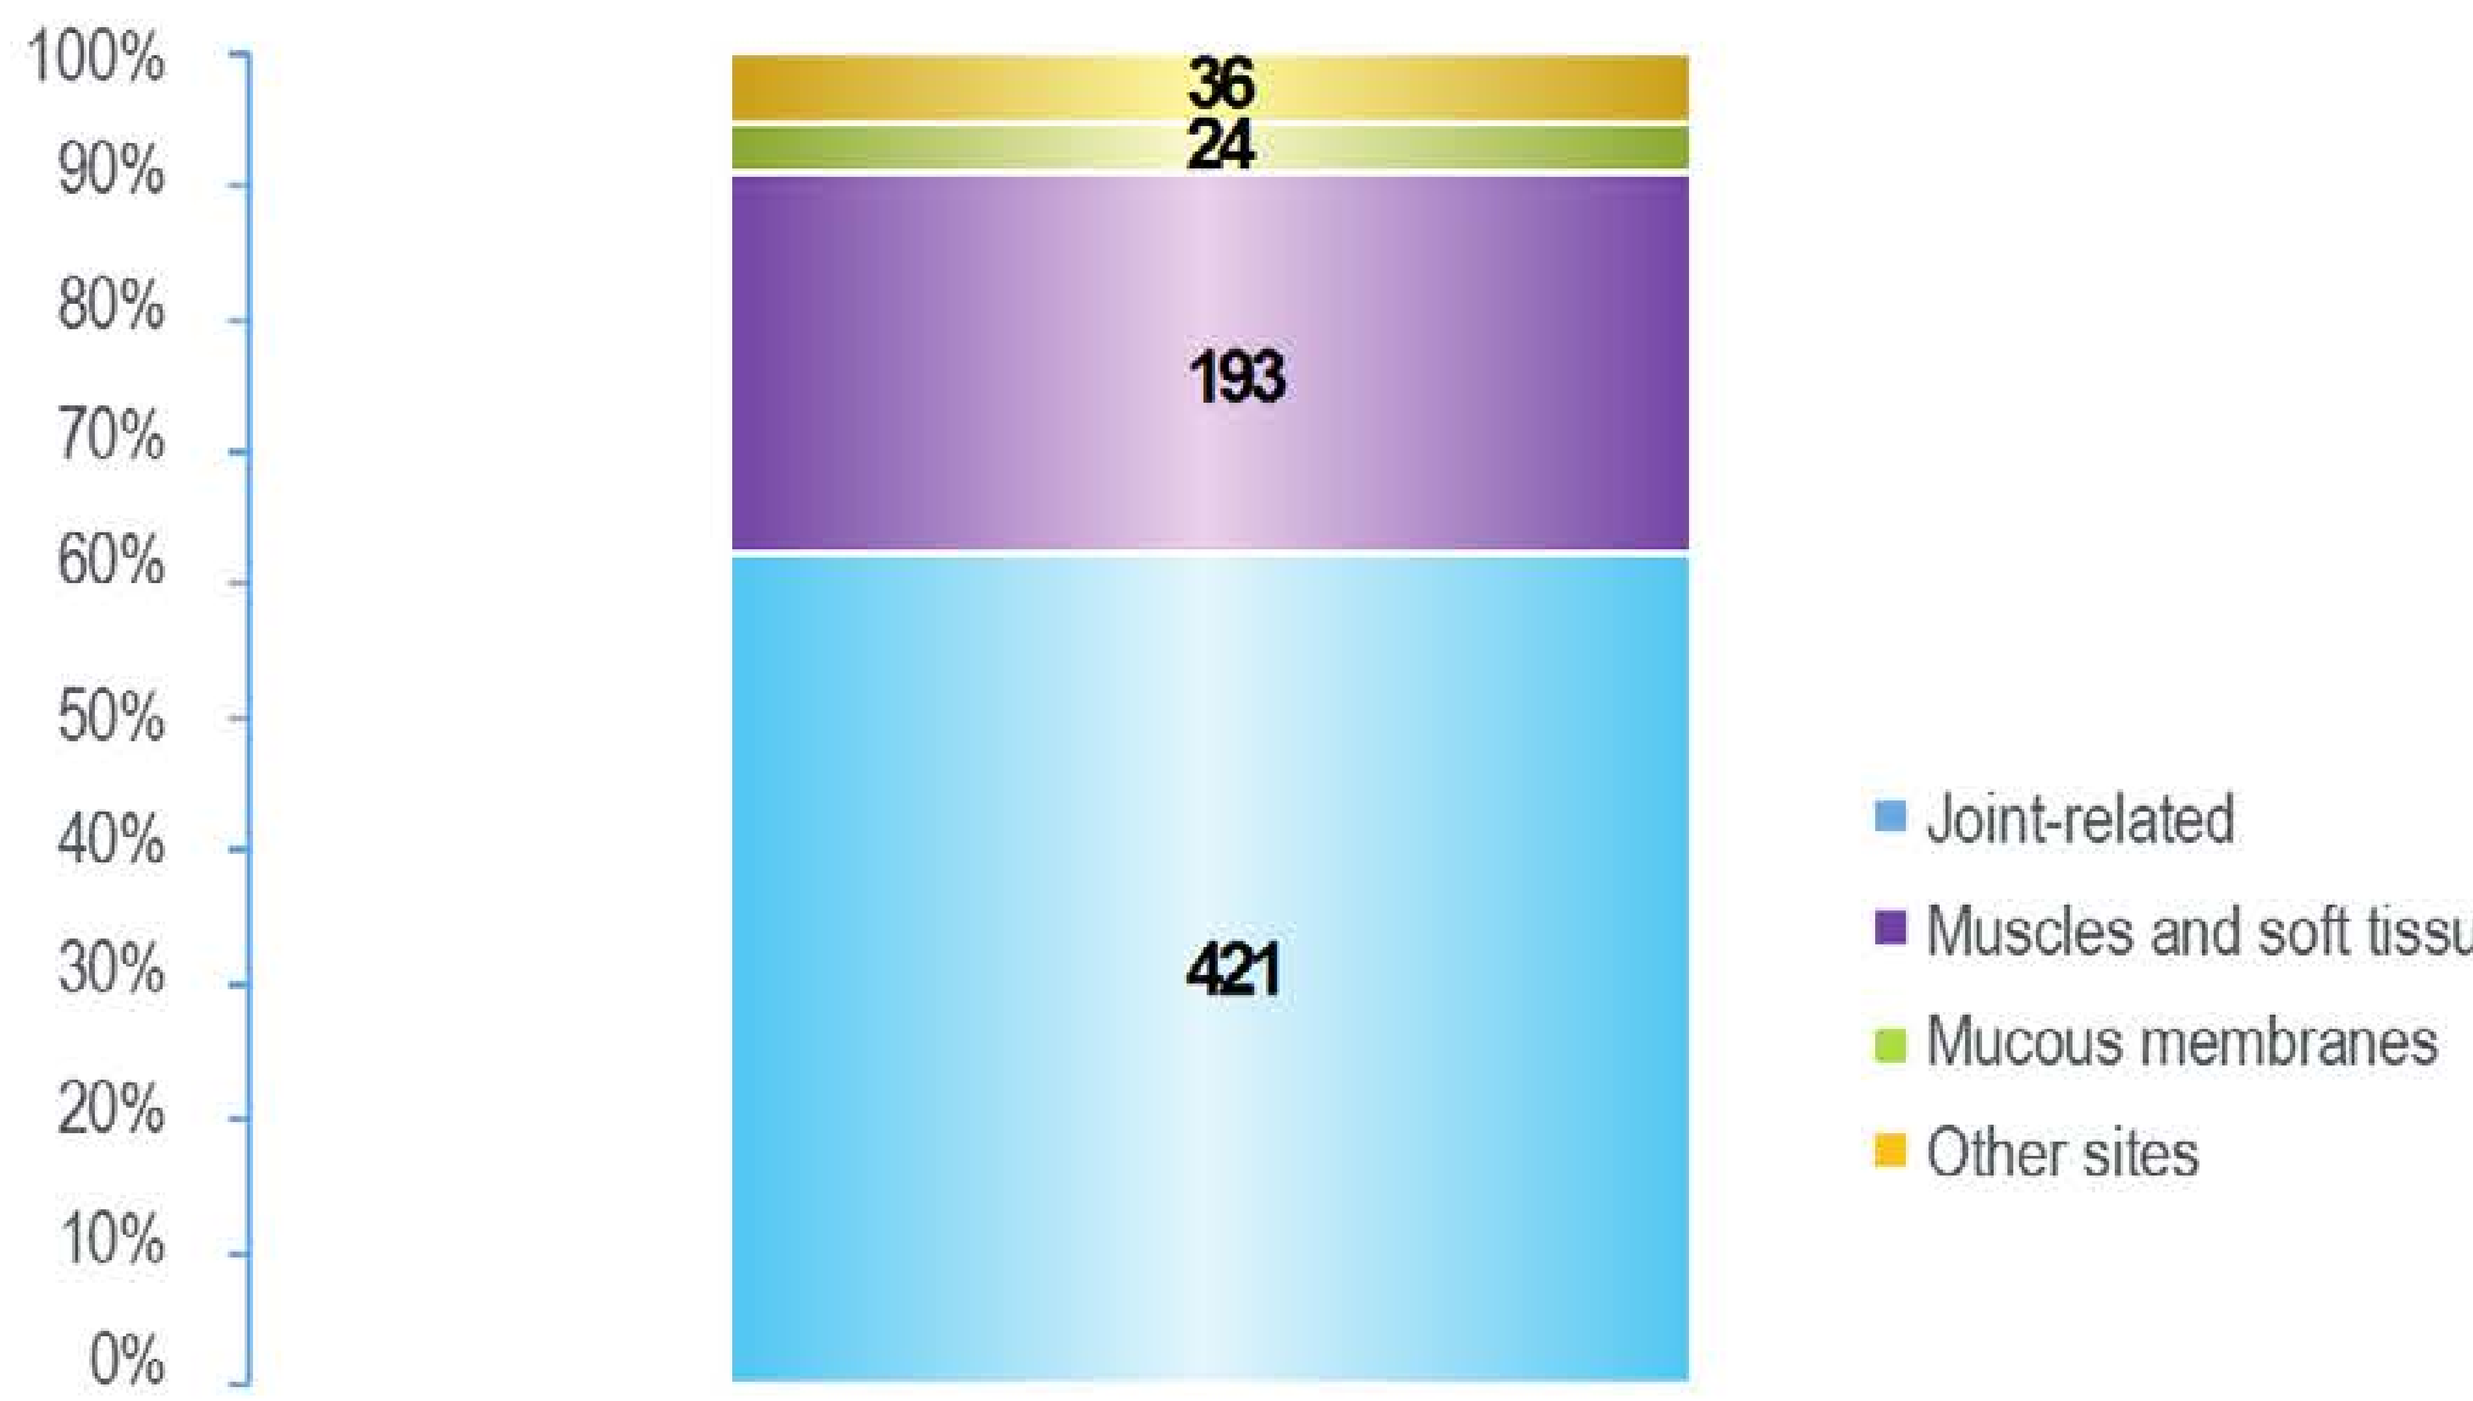
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
36
24
193
421
Joint-related
Muscles and soft tissue
Mucous membranes
Other sites

- The achieved survival was 39.02 LYG for both cohorts (prophylaxis and on-demand).
- The prophylaxis strategy was associated with a higher benefit in QALYs compared to on-demand
- (33.82 vs 22.20). Difference between both strategies was 11.63 QALYs.
- Evolution of bleeds frequency during the 40-year simulation for the cohort of patients treated on-demand is shown in Figure 3 and for the cohort of patients in prophylaxis is shown Figure 4.

Figure 3. Bleeds frequency: patients treated on-demand over time

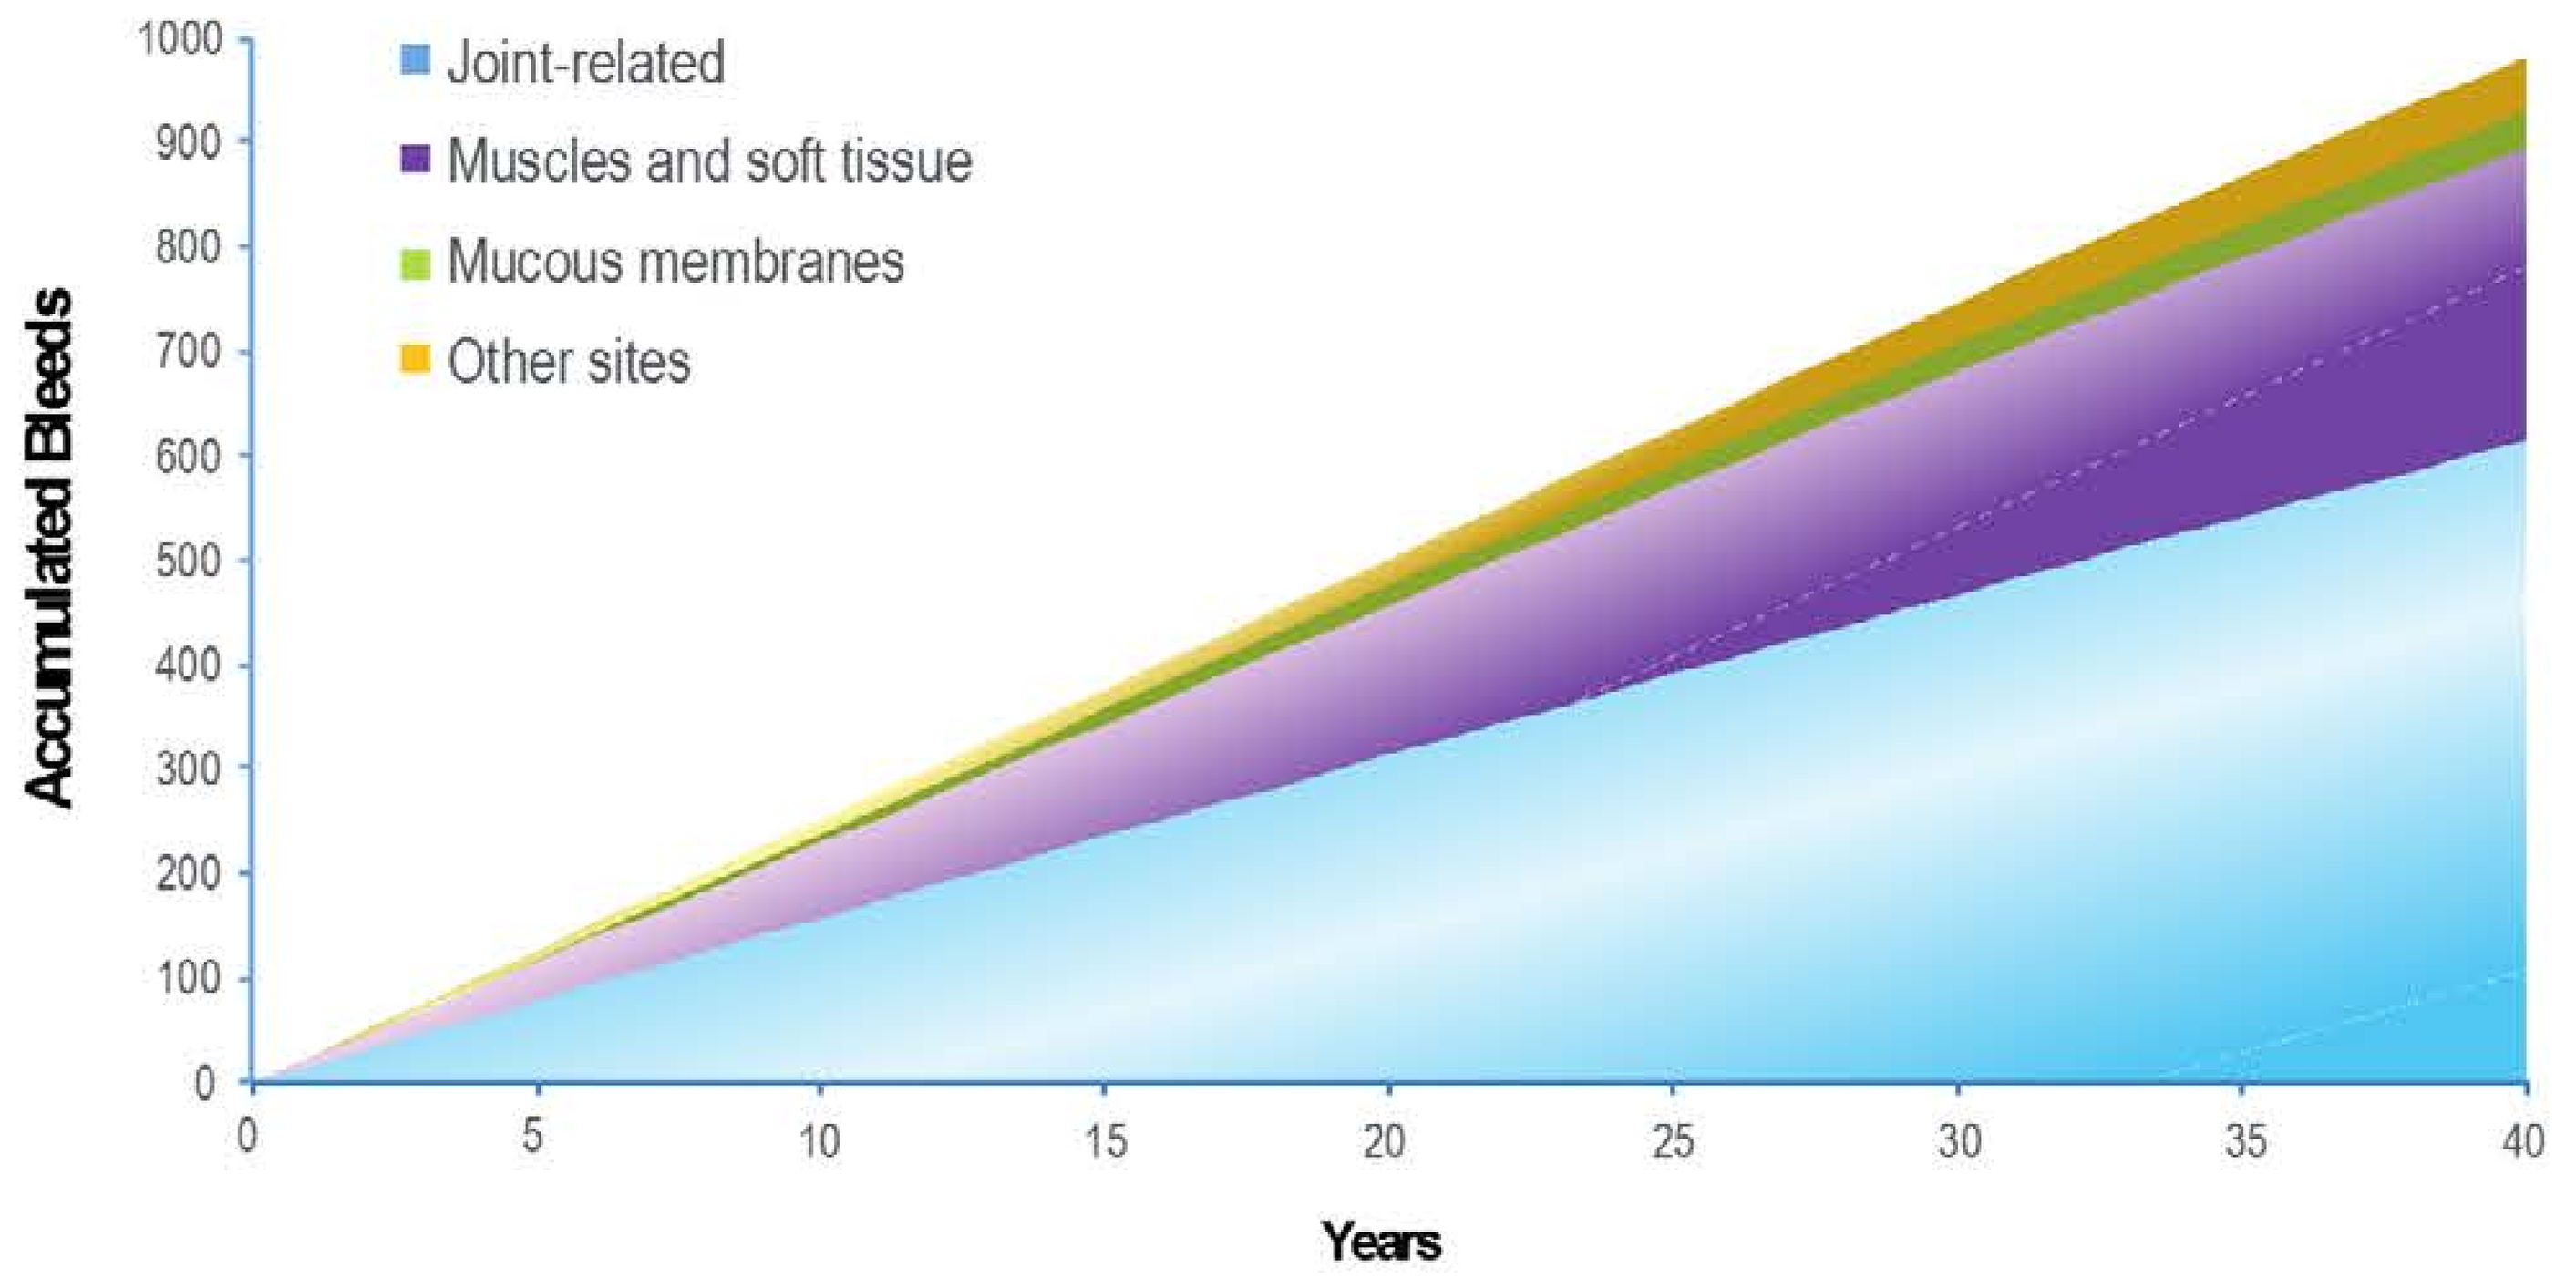
Accumulated Bleeds
1000
900
800
700
600
500
400
300
200
100
0
Joint-related
Muscles and soft tissue
Mucous membranes
Other sites
0
5
10
15
20
25
30
35
40
Years

Figure 4. Bleeds frequency: patients treated with prophylaxis over time

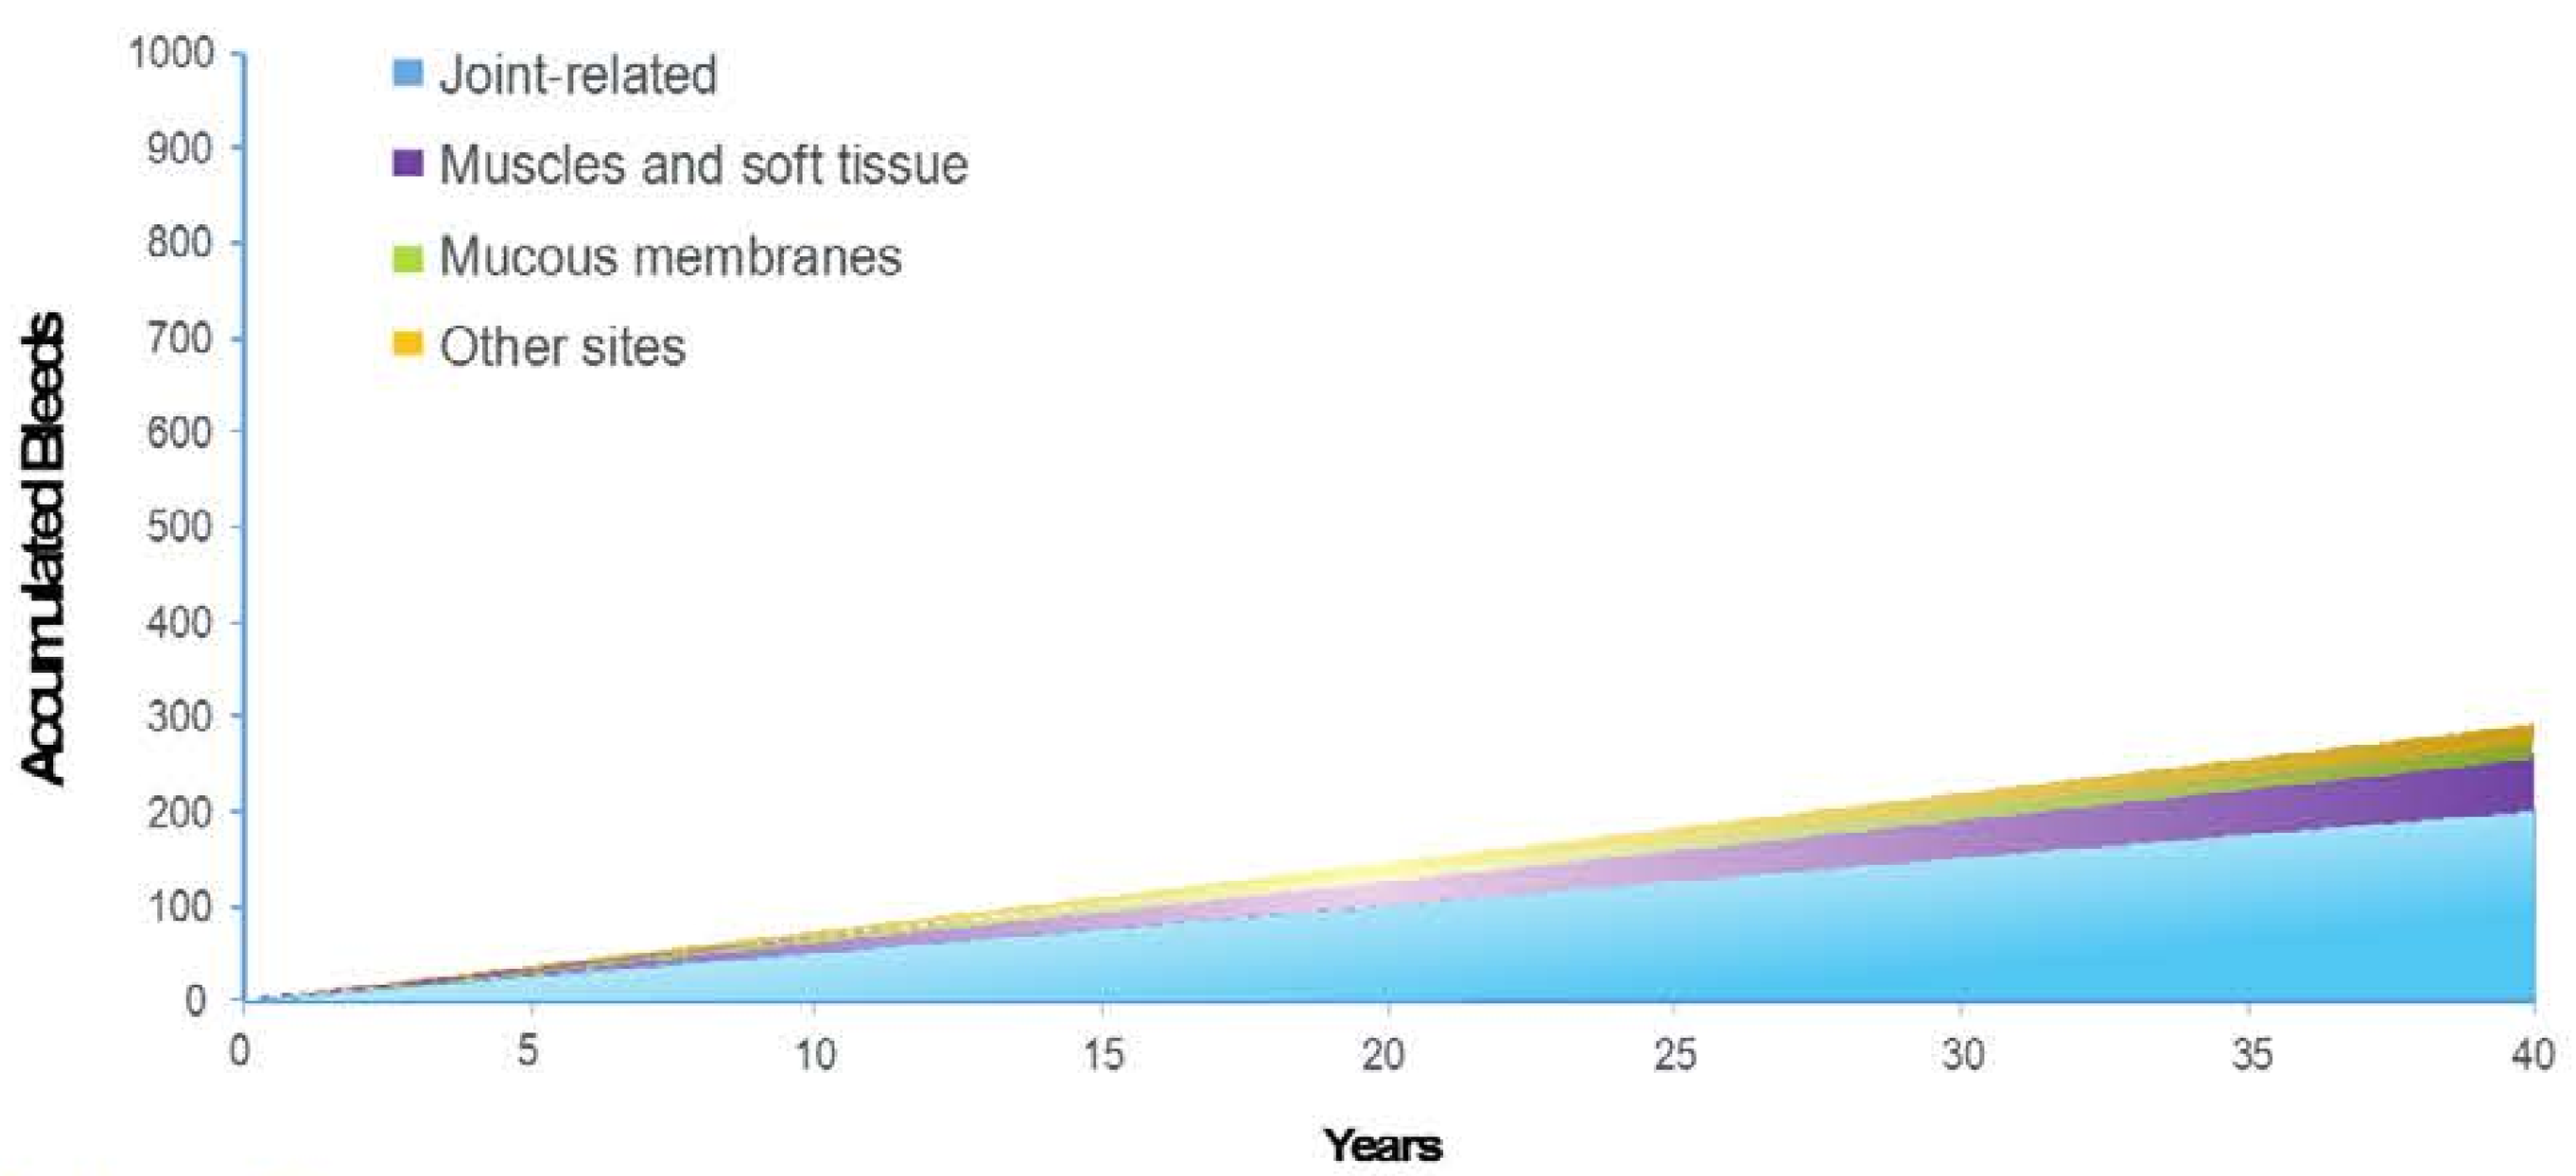
Accumulated Bleeds
1000
900
800
700
600
500
400
300
200
100
0
Joint-related
Muscles and soft tissue
Mucous membranes
Other sites
0
5
10
15
20
25
30
35
40
Years

PSA analysis results

- From the 10,000 Monte-Carlo simulations, the median of bleeds avoided with prophilaxis strategy vs on-demand was 691 (95% Crl; 535-851) in 40 years. Avoided bleeds of prophylaxis strategy vs on-demand evolution during the 40 years of simulations is represented in Figure 5.
- Median of incremental QALYs gained with prophylaxis strategy vs on-demand was 11.65 QALYs (95% Crl; 3.79-19.6).

Figure 5. PSA of bleeds avoided (propylaxis vs on-demand) over time

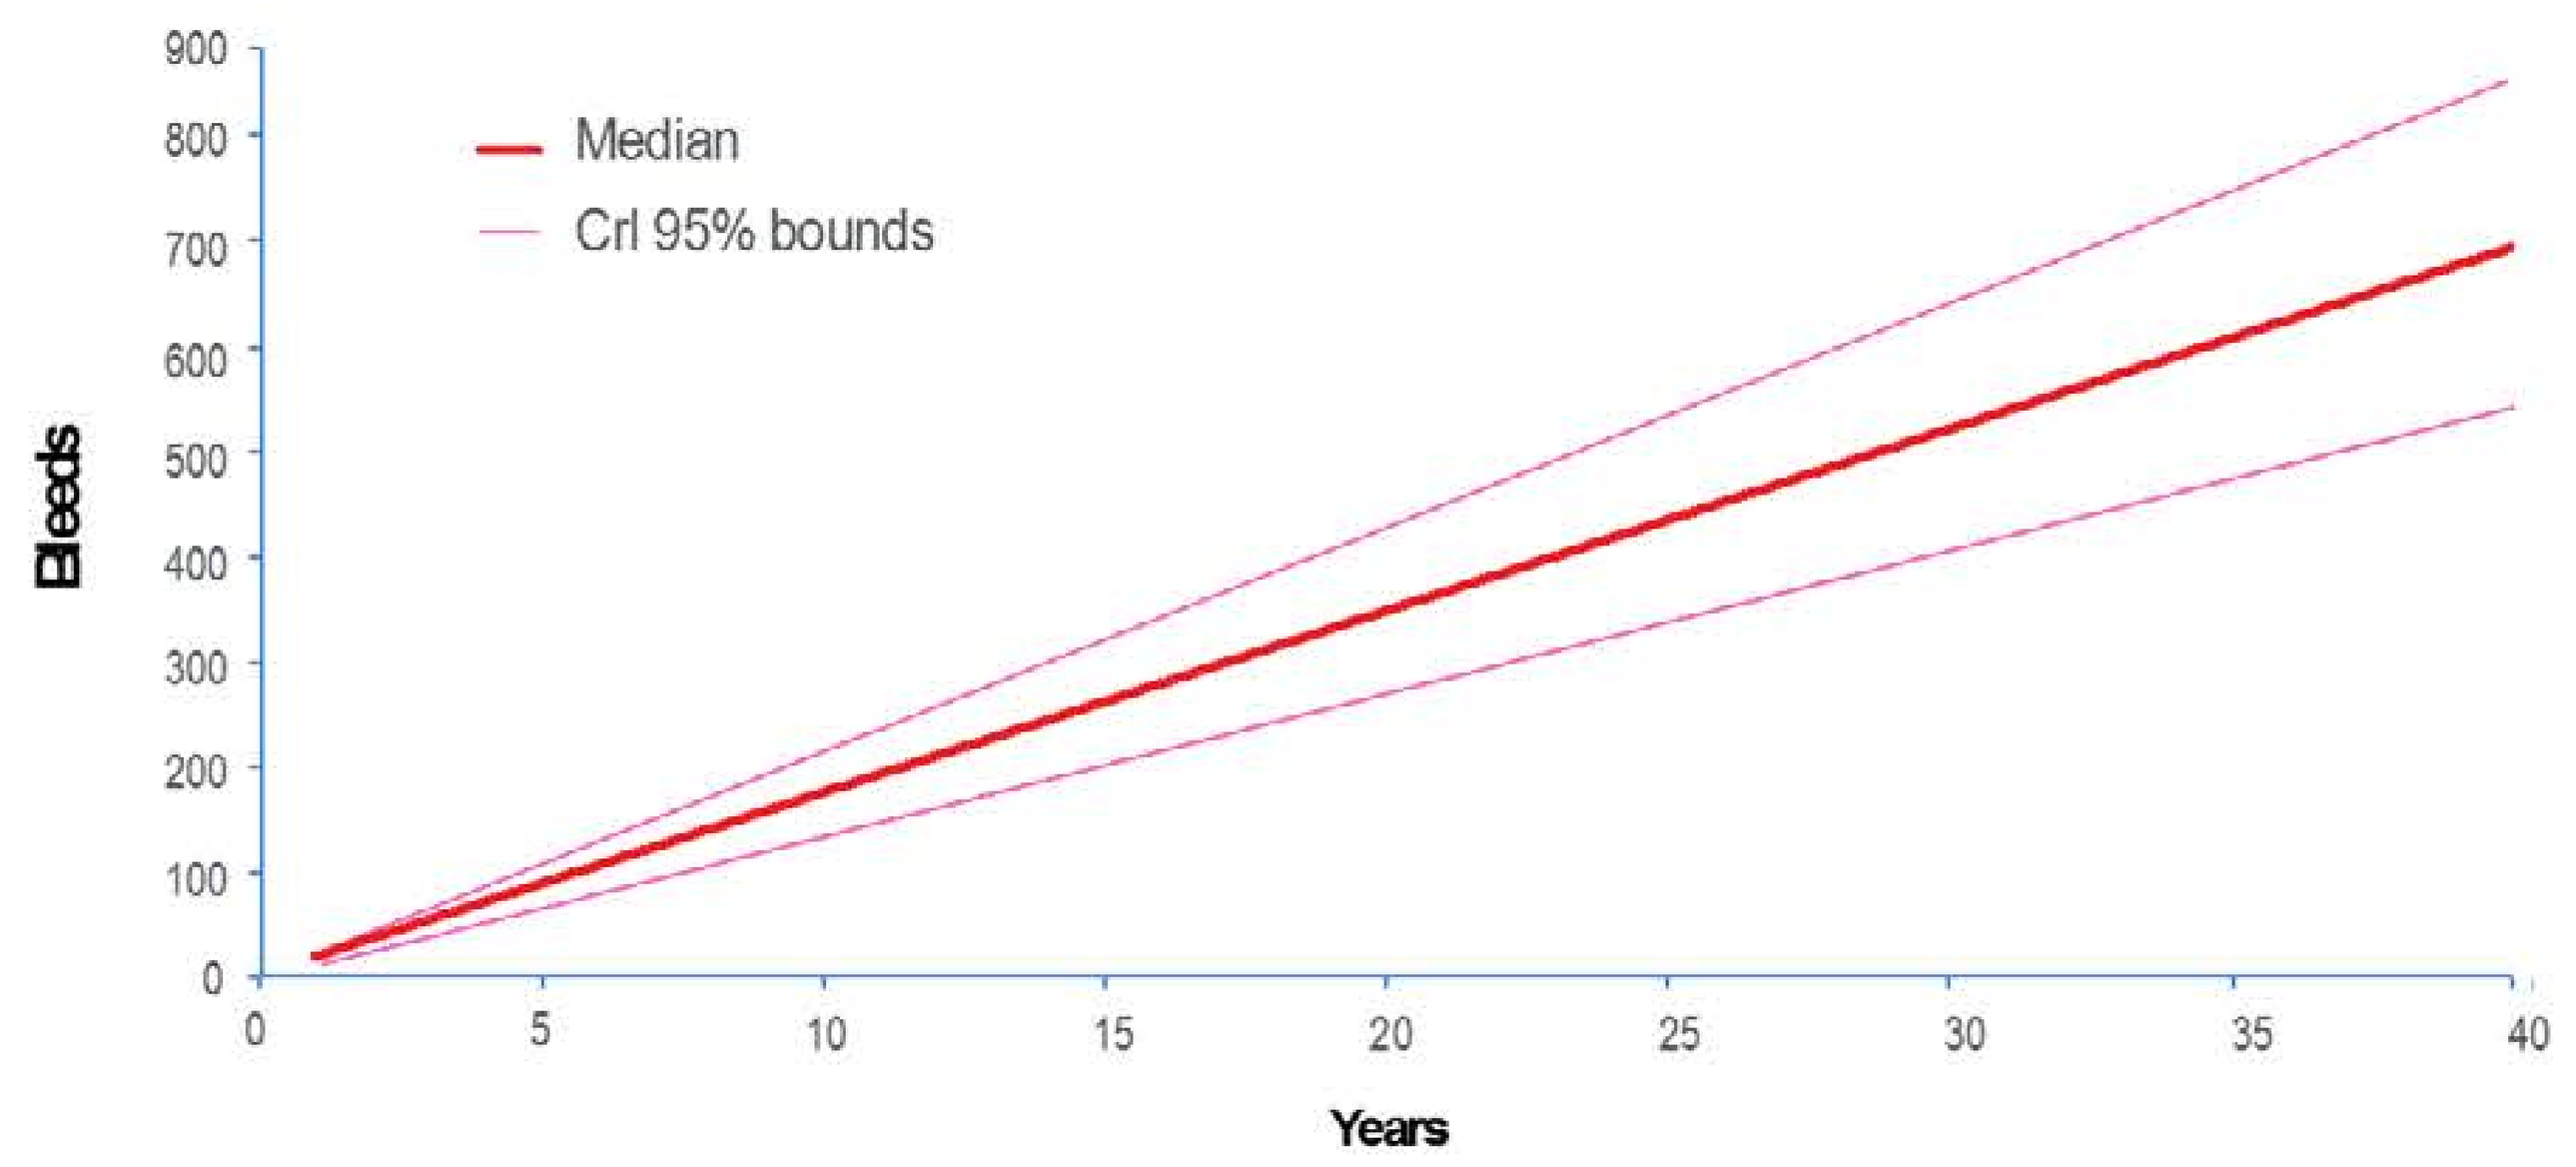
Bleeds
900
800
700
600
500
400
300
200
100
0
Median
Crl 95% bounds
0
5
10
15
20
25
30
35
40
Years

CONCLUSION

- The results of the described modelling approach suggest that prophylaxis treatment regimen with aPCC would be associated with better long-term health outcomes as well as with a significant reduction in the number of bleeding events compared to on-demand treatment in patients with severe haemophilia A with inhibitors

REFERENCES

1. Dimichele D. Inhibitors in Hemophilia: A Primer (Revised Edition). World Federation of Hemophilia (WFH) 2008.
2. Kasper CK. Diagnosis and management of inhibitors to Factors VIII and IX: An Introductory Discussion for Physicians © World Federation of Hemophilia, 2004.
3. Hay CR, Brown S, Collins PW, Keeling DM, Liesner R. The diagnosis and management of factor VIII and IX inhibitors: a guideline from the United Kingdom Haemophilia Centre Doctors Organisation. Br J Haematol. Jun 2006;133(6):591-605.
4. Berntorp E, Shapiro A, Astermark J, et al. Inhibitor treatment in haemophilias A and B: summary statement for the 2006 international consensus conference. Haemophilia. Dec 2006;12 Suppl 6:1-7.
5. Acharya SS, DiMichele DM. Management of factor VIII inhibitors. Best Pract Res Clin Haematol. 2006;19(1):51-66.
6. Hoffman M, Dargaud Y. Mechanisms and monitoring of bypassing agent therapy. J Thromb Haemost. Aug 2012;10(8):1478-1485.
7. Cheng S, et al. FEIBA prophylaxis in hemophilia A patient with inhibitors decreases bleeding episodes, improves joint function and enhance quality of life. Haemophilia 2006;12:Abstract 14 PO 371.
8. Dimichele D, Négrier C. A retrospective postlicensure survey of FEIBA efficacy and safety. Haemophilia. 2006;12:352-62.
9. Lambert T, et al. Can long-term prophylaxis with APCC improve the bleeding rate and quality of life of frequently bleeding haemophiliacs with inhibitor? Haemophilia 2006;12: Abstract 14 PO 399.
10. Leissinger CA, et al. Prophylactic treatment with activated prothrombin complex concentrate (FEIBA) reduces the frequency of bleeding episodes in paediatric patients with haemophilia A and inhibitors. Haemophilia. 2007;13(3):249-55.
11. Valentino LA. Assessing the benefits of FEIBA prophylaxis in haemophilia patients with inhibitors. Haemophilia. 2010;16:263-71.
12. Leissinger C, et al. Anti-Inhibitor Coagulant Complex Prophylaxis in Hemophilia with Inhibitors. N Engl J Med. 2011;365:1684-92.
13. Aznar JA, et al. Haemophilia in Spain. Haemophilia. 2009; 15:665-75.
14. Young G, et al. PRO-PACT: Retrospective observational study on the prophylactic use of recombinant factor VIIa in hemophilia patients with inhibitors. Thromb Res. 2012;130:864-70.
15. Miners H, et al. Cost-Utility analysis of primary prophylaxis versus treatment on-demand for individuals with severe haemophilia. Pharmacoeconomics 2002;20(11):759-774.
16. Instituto Nacional de Estadística. Tablas de mortalidad de la población de España 1991-2013.
17. Miners AH, et al. Assessing health related quality-of-life in individuals with haemophilia. Haemophilia 1999;5:378-85.
18. Naraine VS, et al. Health-related quality-of-life treatments for severe haemophilia: utility measurements using the Standard Gamble technique. Haemophilia. 2002;8(2):112-20.
19. Laupacis A, et al. The effect of elective total hip replacement on health-related quality of life. J Bone Joint Surg Am 1993;75(11): 1619-26.

DISCLOSURES

* Author formerly an employee of Baxalta (5 Baxalta, Madrid, Spain), now part of Shire.

WFH 2016 WORLD CONGRESS
ORLANDO, USA • JULY 24-28, 2016

This study was funded by Shire

Designed by [porib]
pharmacoeconomics & outcomes research iberia

PosterSessionOnline.com

Poster Presented at:

DOI: 10.3252/pso.eu.WFH2016.2016

Health Technology Assessment
Eliazar Sabater

71-PO-T

WFH2016