Reliability and validity of the Haemophilia Activity List (HAL) in von Willebrand disease

K.P.M. van Galen[1] MA Timmer[1] P. de Kleijn[1] K. Fischer[1] W. Foppen[2] R.E.G. Schutgens[1] J. Eikenboom[3] M.H. Cnossen[4] K. Fijnvandraat[5] J.G. van der Bom[3] J. de Meris[6] B.A.P. Laros-van Gorkom[7] K. Meijer[8] F.W.G. Leebeek E.P.[4] Mauser-Bunschoten[1] for the WiN study group

[1]Van Creveldkliniek, University Medical Center Utrecht [2]Radiology, University Medical Center Utrecht [3]Leiden University Medical Center [4]Erasmus University Medical Center, Rotterdam [5]Academic Medical Center Amsterdam [6]Dutch patient society on haemophilia [7]Radbout university medical center, Nijmegen, [8]University Medical Center Groningen, The Netherlands

Aim

To validate the Haemophilia Activity List (HAL) in Von Willebrand disease.

Background

Haemophilia arthropathy due to joint bleeds occurs in VWD patients. To examine functional limitations due to joint bleeds the Haemophilia activity list (HAL) questionnaire has been developed in haemophilia. The performance and feasibility of this instrument in VWD patients is currently unknown.

Methods

- Reliability: internal consistency of the 7 domains and 3 components of the HAL (Cronbach's α>0.7 acceptable).
- Construct validity: correlation of the Figure 8 walking test on maximum speed with the HAL-subscore legs function (Spearman's *r* (r_s)>0.60 acceptable).
- Construct validity: we hypothesized that the total HAL score should be lower in patients with type 3 VWD (VWF<5%) and in patients with radiologic joint damage on X ray (p<0.10 Mann Whitney U)
- Construct validity: correlation of the HAL with the physical component domains of the SF36 questionnaire and with the sum score on the 5 domains of the Impact on Participation and Autonomy (IPA) questionnaire r_s>0.60 acceptable).

Score		Items
Lying / sitting / kneeling / standing	LSKS	1-8 (8)
Functions of the legs	LEGS	9-17 (9)
Functions of the arms	ARMS	18-21 (4)
Use of transportation	TRANS	22-24 (3)
Self care	SELFC	25-29 (5)
Household tasks	HOUSEH	30-35 (6)
Leisure activities and sports	LEISPO	36-42 (7)
Upper Extremity Activities	UPPER	* (9)
Basic Lower Extremity Activities	LOWBAS	** (6)
Complex Lower Extremity Activities	LOWCOM	*** (9)
Sum score	SUM	1-42 (42)

* Items for UPPER-component: 18, 19, 20, 21, 25, 26, 27, 28, 29. (9 items)
** Items for LOWBAS-component: 8, 9, 10, 11, 12, 13. (6 items)
*** Items for LOWCOM-component: 3, 4, 6, 7, 14, 15, 16, 17, 22. (9 items)

Figure 1: HAL domains

Study population

- 95 adult VWD patients with historically lowest VWF levels ≤30 U/dl and/or FVIII:C ≤40 U/dl and a bleeding- or family history of VWD; type 3 VWD defined as vWF activity<5 U/dL
- 60% male, median age 47 years
- Mean VWF activity 11 U/dL and FVIII:C 28 U/dL
- 48 patients (51%) had a documented history of joint bleeds

Results

The HAL questionnaire was completed by 95 patients with VWD. The total score ranged from 26.5 to 100. 23/47 patients without a history of joint bleeds reached the ceiling score of 100, compared to 10/48 patients with joint bleeds (p=0.004). Internal consistency was good for the 7 subdomains (α=0.90) and 3 components (α=0.89). The HAL subscore legs function correlated with the Figure 8 test (r_s=0.61, n=94). The total HAL score also correlated with the SF36 subdomain 'physical functioning' (r_s=0.65, n=73), the physical component summary score (r_s=0.62, n=70) and with the sum score of the 5 IPA domains (r_s=0.69, n=76). Type 3 VWD and VWD patients with radiological joint damage had significantly lower HAL scores compared to the other VWD patients (Table 1).

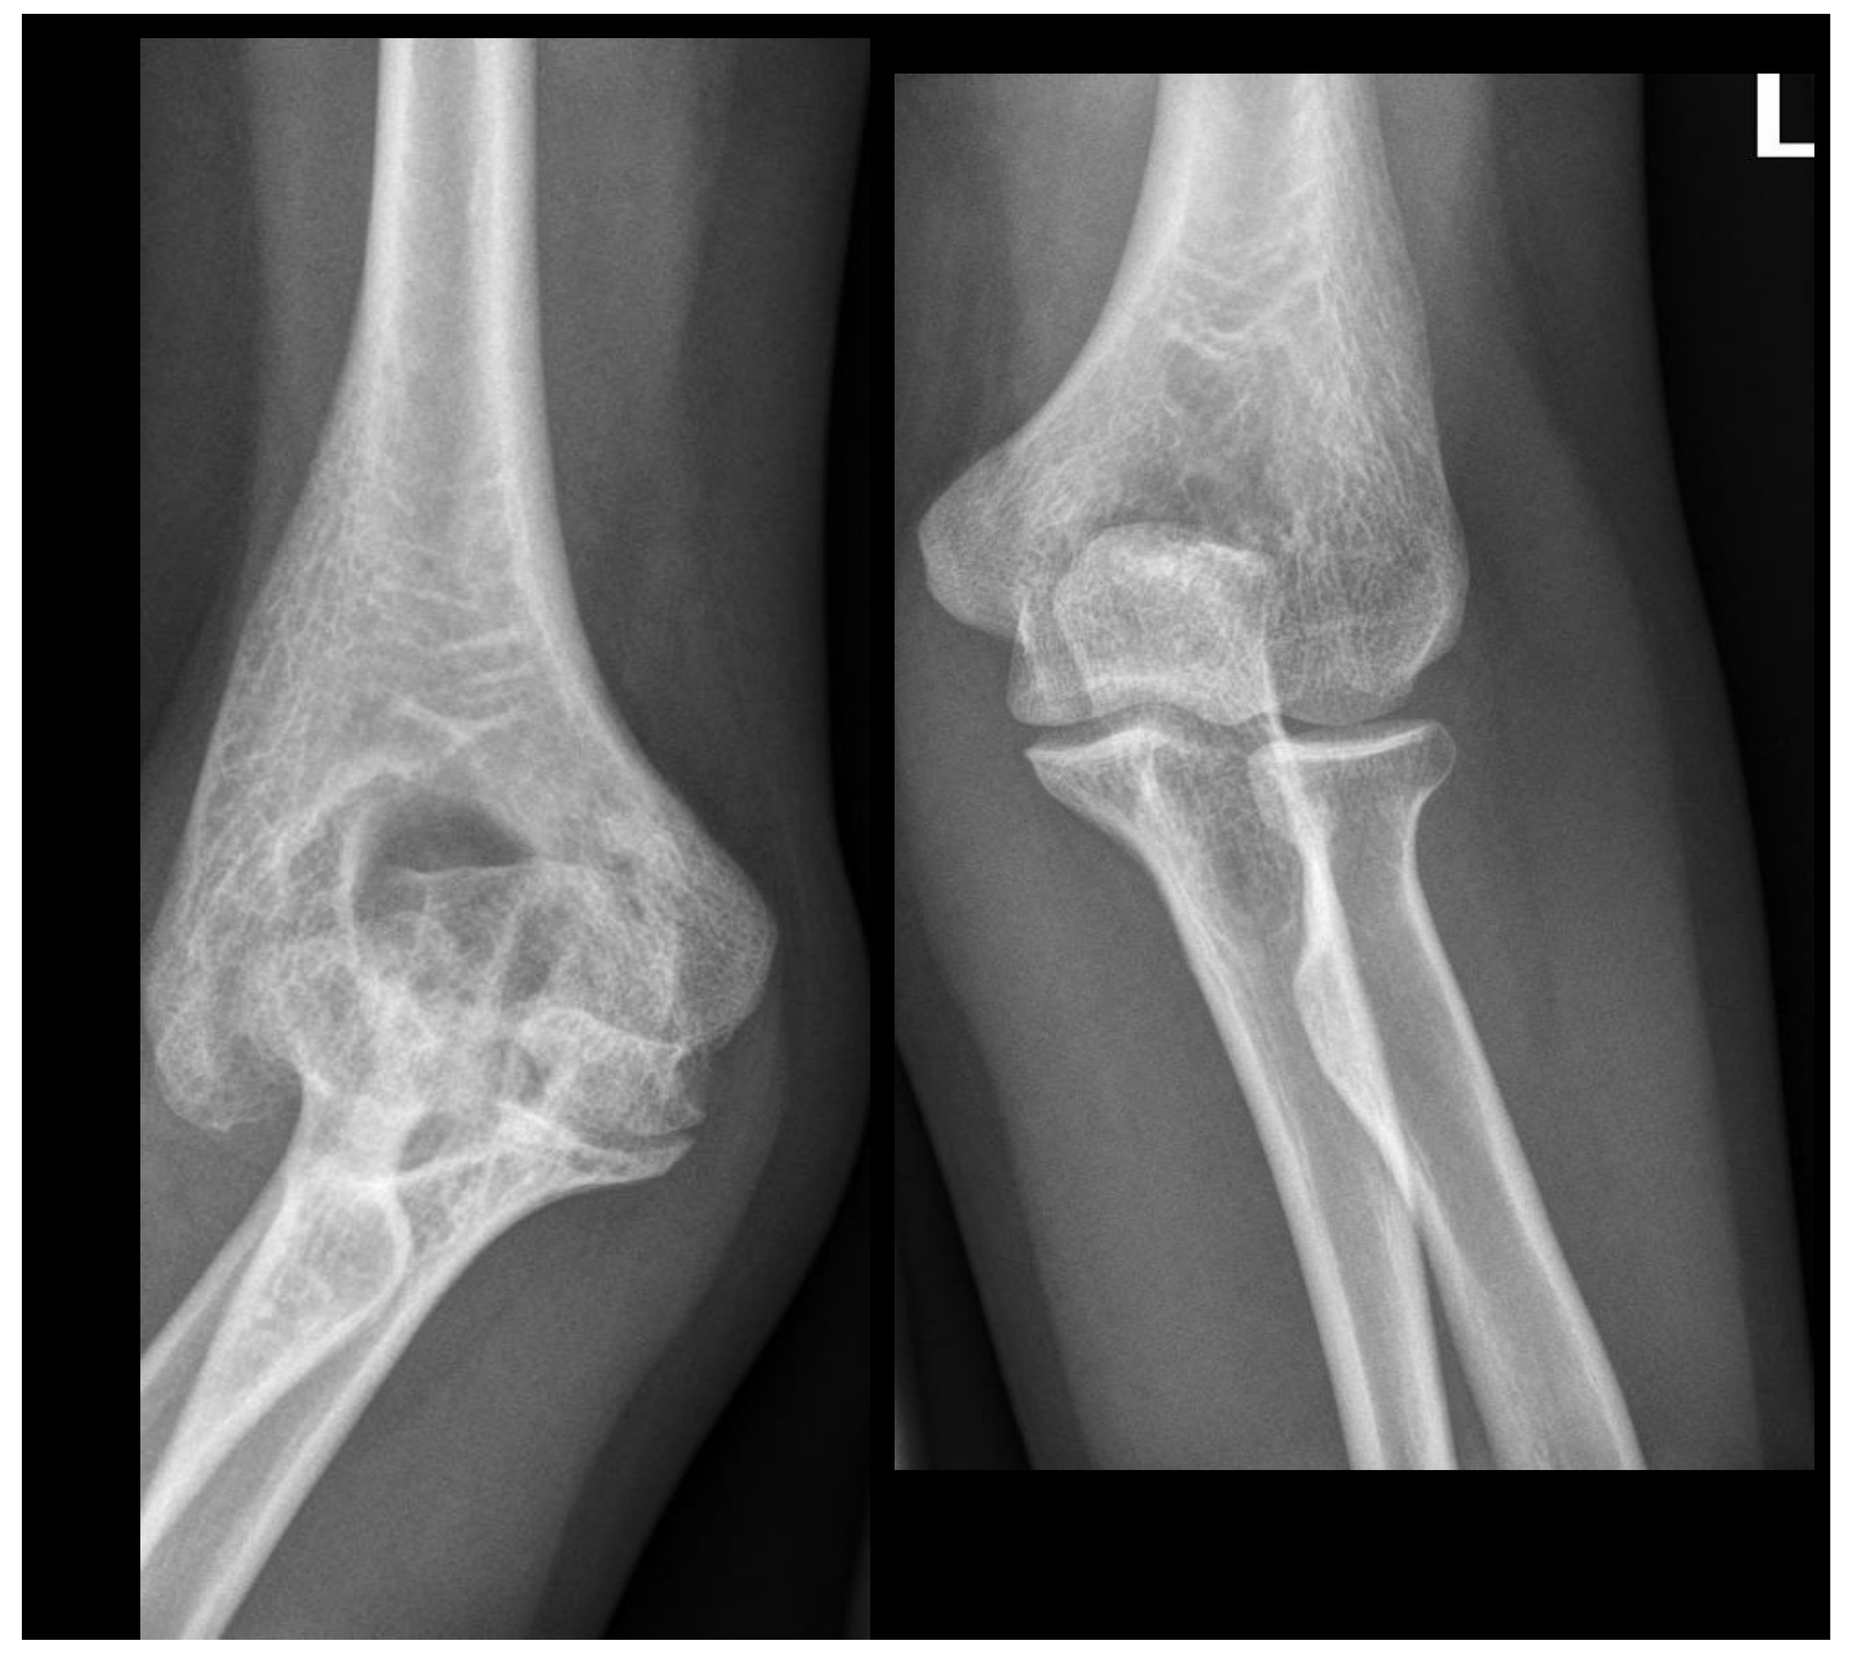
L

Figure 2: Radiologic joint damage of the right elbow in a patient with type 3 VWD (Pettersson score 11, range 0-13 per joint)

Table 1: construct validity of the HAL in VWD

	Type 3 VWD n=23	Non-type 3 VWD n=72	VWD patients with radiologic joint damage* n=50	VWD patients without radiologic joint damage n=42
HAL total score (median, IQR)	78 (66-95)**	97 (83-100)	89 (68-100)**	99 (91-100)

* X-ray Pettersson score (PS) > 0
** p<0.01 compared to non type 3 VWD patients and those with PS 0

Conclusion

Considering the maximum HAL score as no functional limitations, the HAL appears sufficiently reliable and valid to assess functional limitations in VWD patients with a history of joint bleeds.

Van Creveldkliniek, University Medical Center Utrecht, Utrecht, the Netherlands
Correspondence: k.p.m.vangalen@umcutrecht.nl; m.a.timmer@umcutrecht.nl

The present study was funded by an unrestricted grant from CSL Behring and the Dutch Haemophilia Foundation

PosterSessionOnline.com
Poster Presented at:
DOI: 10.3252/pso.eu.WFH2016.2016
Von Willebrand disease
Karin van
193--PO-T
WFH2016