The Development of the World Bleeding Disorder Registry Pilot Phase

J O'Hara[1], S Diop[2], R Hollingsworth[3], A Srivastava[4], D Lillicrap[5], HM van den Berg[6], C Herr[7], D Coffin[7], A Iorio[8], GF Pierce[7]

[1]HCD Economics, London, UK; [2]Cheikh Anta Diop University, Dakar, Senegal; [3]Medical Data Solutions and Services (MDSAS), Manchester, UK; [4]Christian Medical College, Vellore, India; [5]Queen's University, Kingston, Canada; [6]University Medical Center, Utrecht, The Netherlands; [7]World Federation of Hemophilia, Montreal, Canada; [8]McMaster University, Hamilton, Canada

Introduction and Objective

- Management of hemophilia has benefited from country specific and national registries, however many countries do not have a standardized data collection system.
- The World Federation of Hemophilia (WFH) is seeking to develop an international World Bleeding Disorder Registry (WBDR).
- This will help hemophilia treatment centres in assessing patients over time and may improve hemophilia care.
- Prior to implementing the WBDR, a pilot phase is being conducted to assess the feasibility of the project.

Materials and Methods

- HTCs of varying levels of economic development (emerging, mid-level and established) were invited to participate in the pilot phase.
- Each HTC is expected to enrol approximately 10 patients with hemophilia.
- A Universal Case Report Form (U-CRF) was developed based on published recommendations and consensus of world experts in hemophilia.

WFH

WORLD FEDERATION OF HEMOPHILIA
FÉDÉRATION MONDIALE DE L'HÉMOPHILIE
FEDERACIÓN MUNDIAL DE HEMOFILIA

Results

Participating Hemophilia Treatment Centers

40 HTCs invited, 31 participating = **78% Participation Rate**

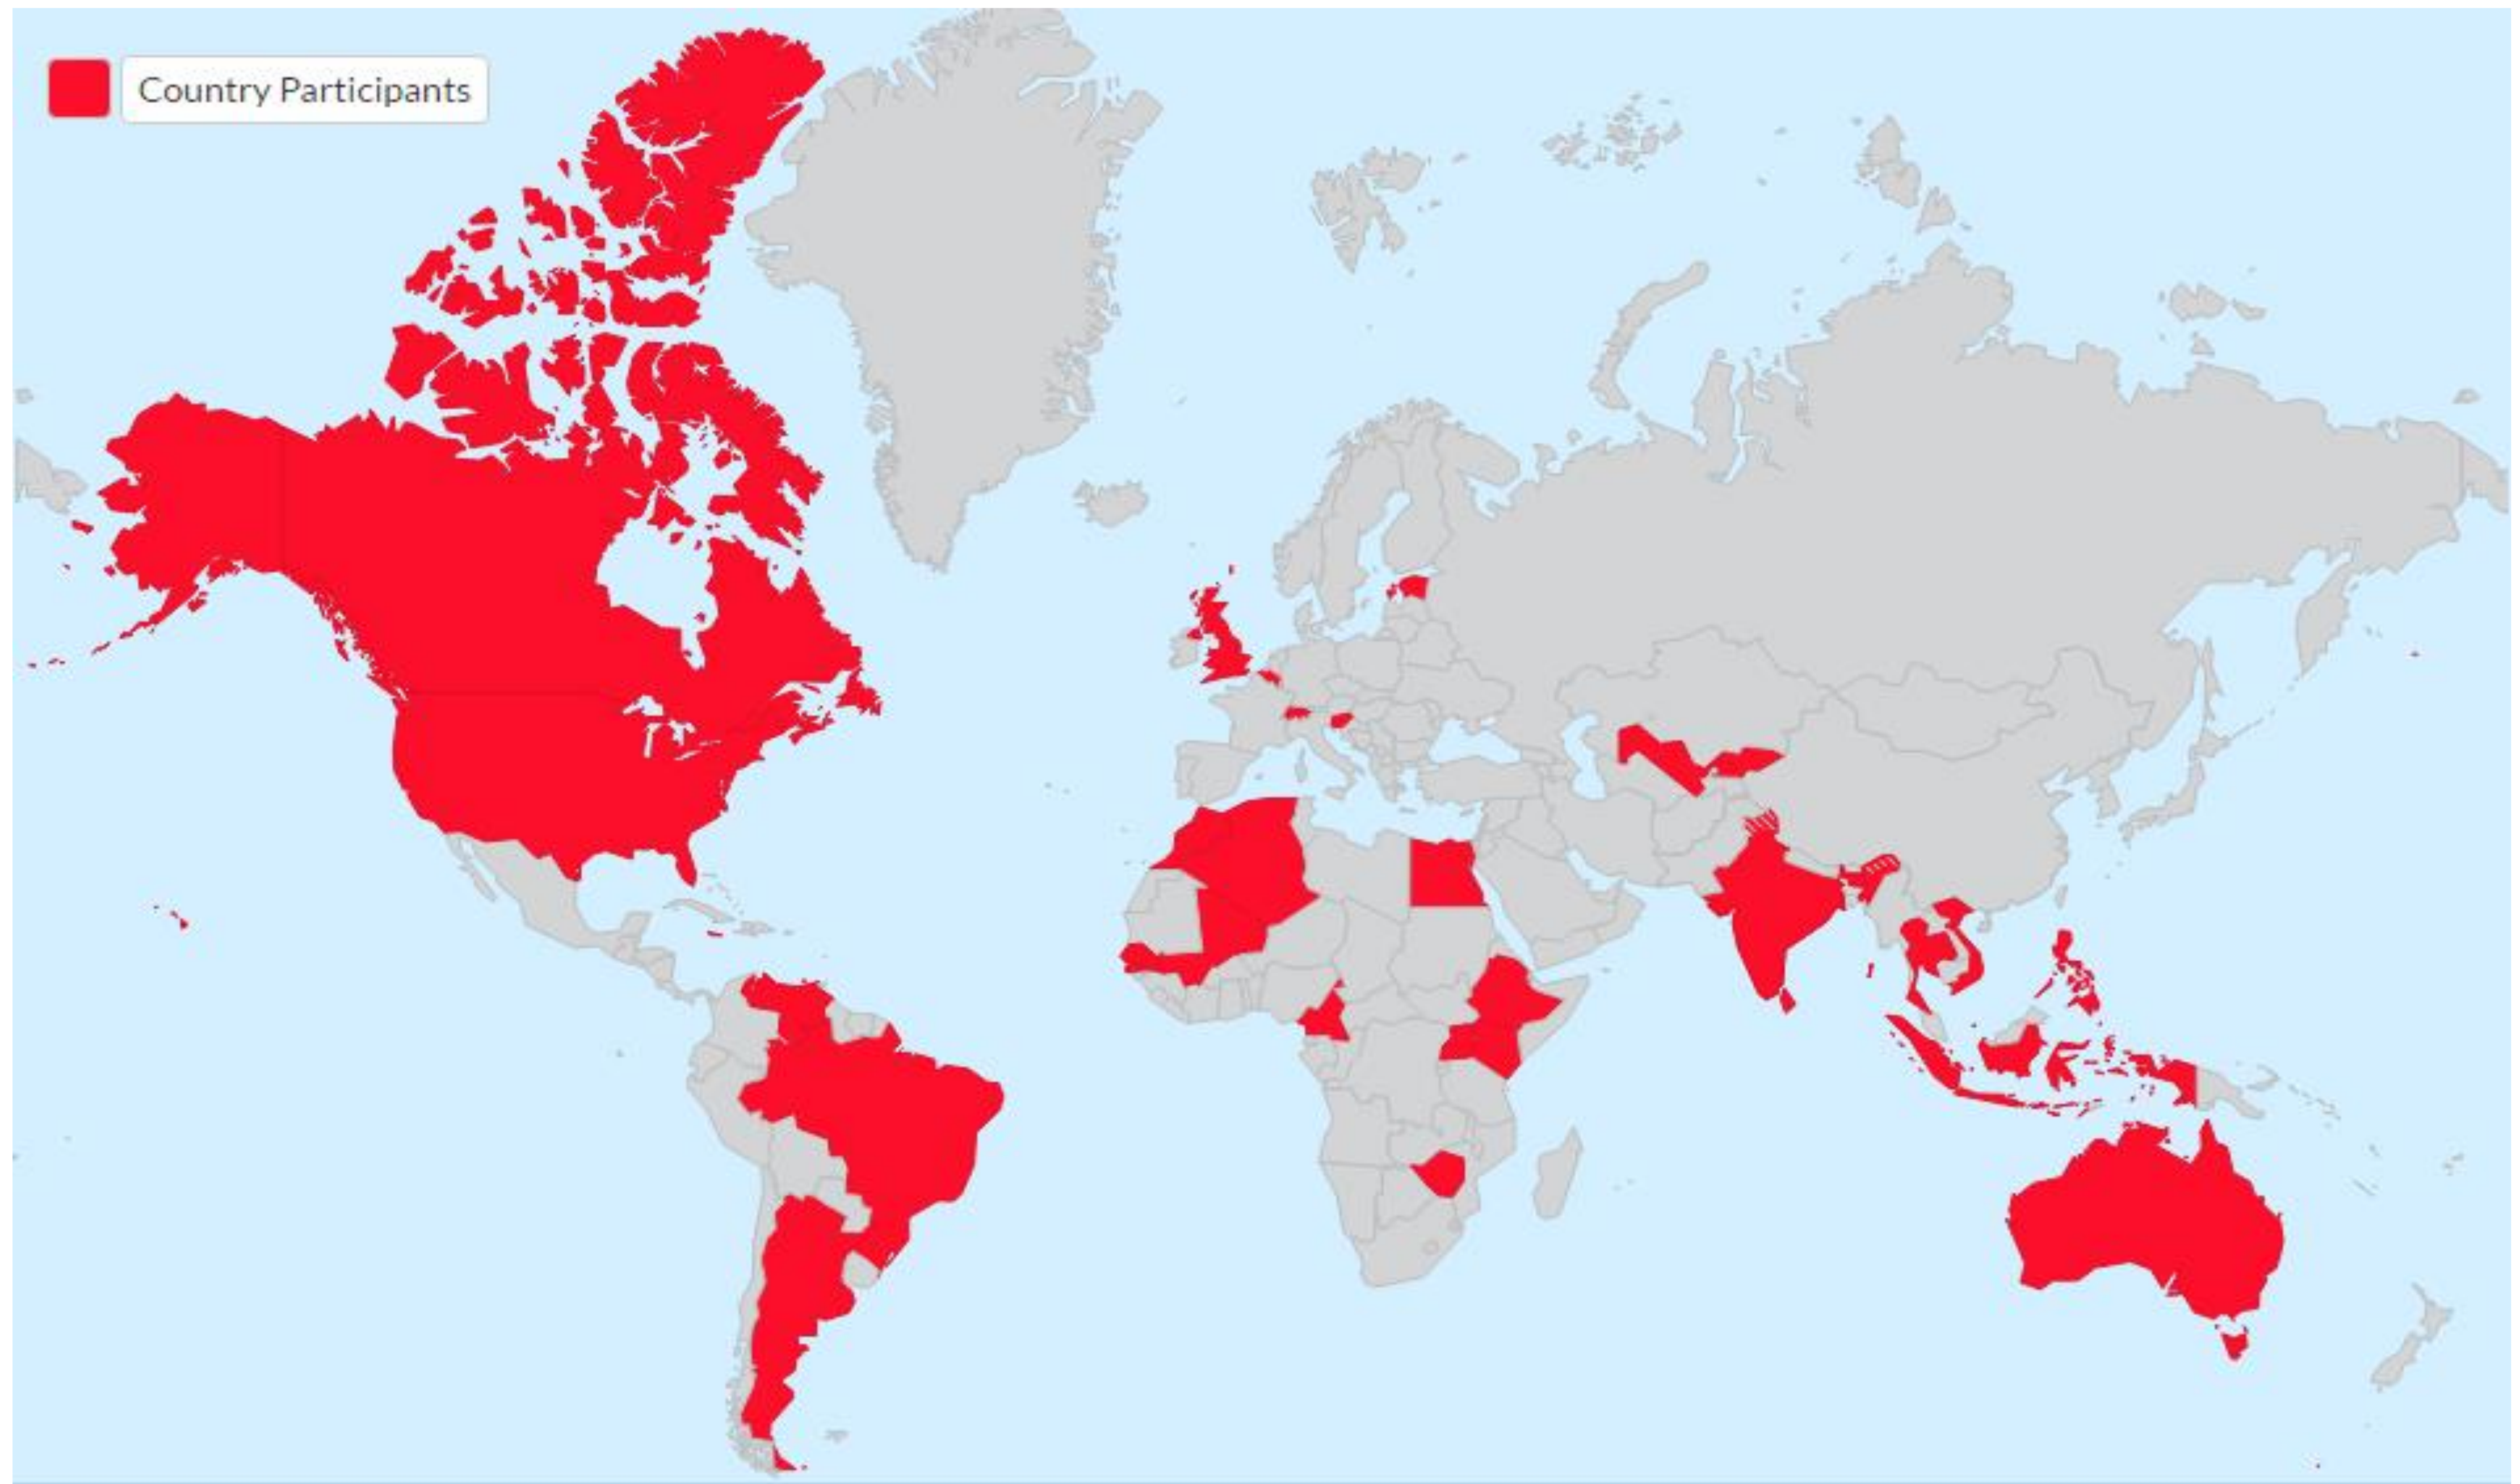
Country Participants

Participating HTCs: Algeria, Argentina, Australia, Belgium, Brazil, Cameroon, Canada, Egypt, Estonia, Ethiopia, India, India, Indonesia, Jamaica, Kenya, Kyrgyzstan, Mali, Morocco, Philippines, Senegal, Slovenia, Sri Lanka, Switzerland, Thailand, Uganda, United Kingdom, United States of America, Uzbekistan, Venezuela, Vietnam, Zimbabwe

Conclusions

- The results of the pilot phase of the WBDR will be reported in Q1 of 2017.
- Engagement from the HTCs has been encouraging, with 78% of invited HTCs having agreed to participate.
- The feasibility of implementing a larger scale WBDR for use by HTCs worldwide will be based on performance indicators and will be assessed in December 2016.

Performance Indicators of Pilot Phase

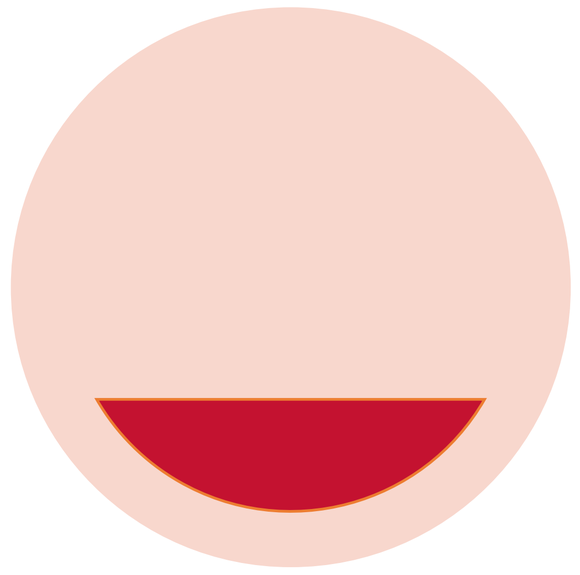

Proportion of HTCs invited who participate;

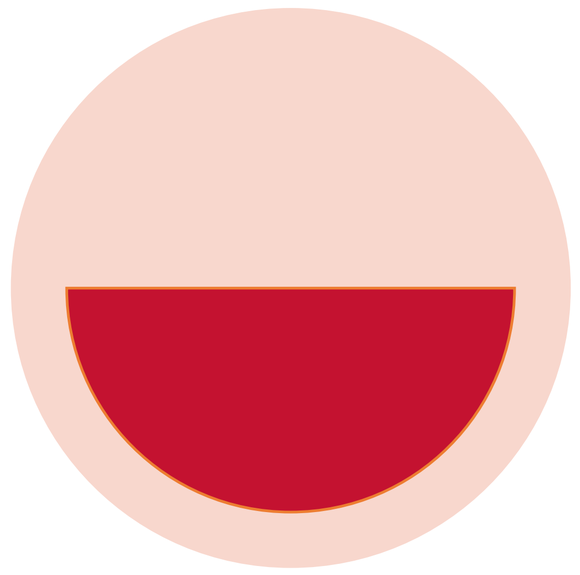

Proportion of participating HTCs who successfully obtain regulatory approval;

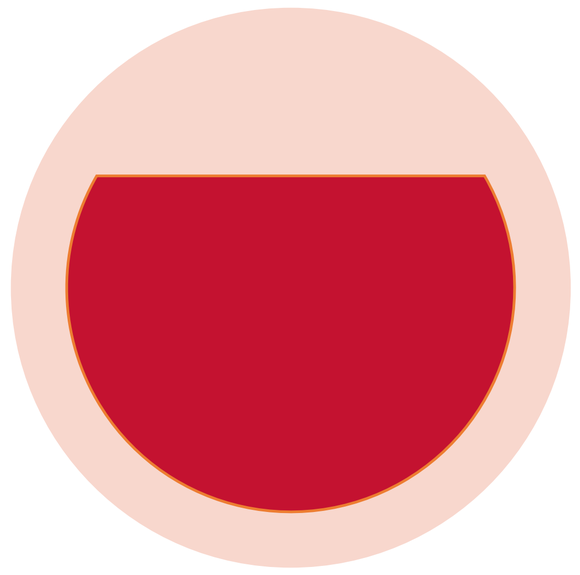

Proportion of patients approached who consent to participate;

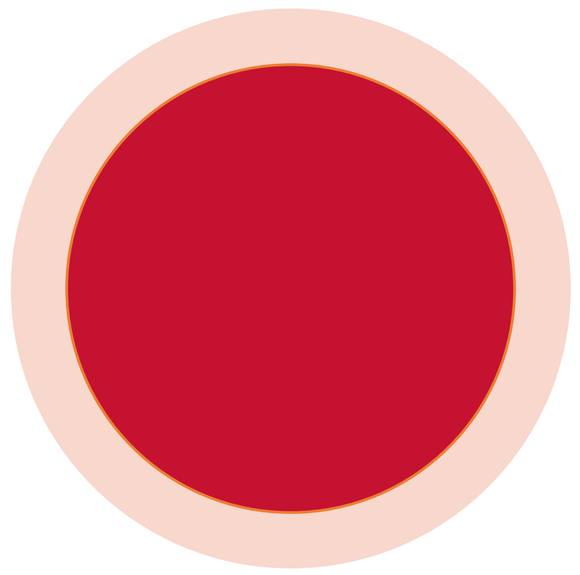

Proportion of enrolled patients on which data collection and database entry is successful.

Universal Case Report Form (U-CRF)

- The U-CRF is available at http://www.wfh.org/en/u-crf for use by the hemophilia community, with the aim of encouraging a standardized approach to documentation of clinical care on patients with hemophilia around the world.

Acknowledgements

- We thank the participating Hemophilia Treatment Centres for their support in the pilot phase of the WBDR.
- *The WFH would like to thank the Hemophilia Center of Western Pennsylvania for its support of the WFH Epidemiological Research Program.*
- The WFH Epidemiological Research Program is supported by funding from Baxalta, Bayer and Grifols.

PosterSessionOnline.com
Poster Presented at: WFH 2016 World Congress
DOI: 10.3252/pso.eu.WFH2016.2016
Data and Demographics
Jamie OHara
2-LB
WFH2016