Immune monitoring of FVIII inhibitor development in the Hemophilia Inhibitor PUP Study (HIPS)

Poster P-T-044

Hofbauer CJ[1], Male C[2], Brown D[3], Santagostino E[4], Oldenburg J[5], Scheiflinger F[1], Reipert BM[1]

[1]Baxter Bioscience, Vienna, Austria, [2]Medical University, Vienna, Austria, [3]University of Texas, Houston, USA, [4]University of Milan, Italy, [5]University of Bonn, Germany

Introduction

The development of neutralizing antibodies (FVIII inhibitors) is the major treatment complication during FVIII substitution therapy of hemophilia A patients. Both clinical and experimental data support the central role of CD4+ T cells in the regulation of FVIII inhibitors [1,2]. However, very little information is available on the evolution of FVIII-specific immune responses in patients during the early phases of FVIII substitution therapy.

FVIII-specific immune monitoring during prospective, longitudinal studies would help to gain further understanding of the interplay between genetic and environmental influences in the evolution of anti-FVIII immune responses and the role of antigen specific CD4+ T cells in this process. This information will facilitate improvement of hemophilia A care and quality of life for patients afflicted from FVIII inhibitors.

Objective

The aim was to establish a comprehensive set of technologies for FVIII-specific immune monitoring in hemophilia A patients during early exposure days to FVIII. This poster illustrates "proof of principle data" collected during method validation. The developed technologies are currently applied in the Hemophilia Inhibitor PUP Study (HIPS).

HIPS is an investigator-initiated, prospective clinical study funded by Baxter Healthcare Cooperation. The primary objective is to monitor biomarkers of FVIII-specific immune responses within the first 50 exposure days to recombinant human FVIII (ADVATE®) in previously untreated patients (PUPs) suffering from severe Hemophilia A. Before first treatment and at regular intervals thereafter peripheral blood samples are be taken for immune monitoring (Figure 1). The study is open for recruitment.

Methods

Ig isotypes and IgG subclasses of FVIII-binding antibodies (Ab)

Multi-step analysis of Ab titers in citrated plasma is done using semi-quantitative ELISA assays for the detection of human FVIII-binding IgM, IgA, IgG1, IgG2, IgG3 and IgG4 [3].

Apparent affinity of FVIII-specific binding antibodies

Apparent affinity of FVIII-binding Ab in citrated plasma is assessed by ELISA based competition assays as described by Bobrovnik et al [4].

FVIII-specific CD4+ T cell signatures

PBMCs are isolated from citrated blood samples and in vitro restimulated with human recombinant FVIII (10µg/ml) for 6 hours. Activation of FVIII-specific memory CD4+ T cell responses is detected via analysis of gene expression patterns using microarray technology (Agilent). Data analysis is done using IPA® software (Ingenuity Systems).

Patients and healthy blood donors

Peripheral blood samples were received after written informed consent from patients with severe hemophilia A and from healthy blood donors with local ethical committee approval.

Figure 1: Early treatment with FVIII products is decisive for FVIII inhibitor development

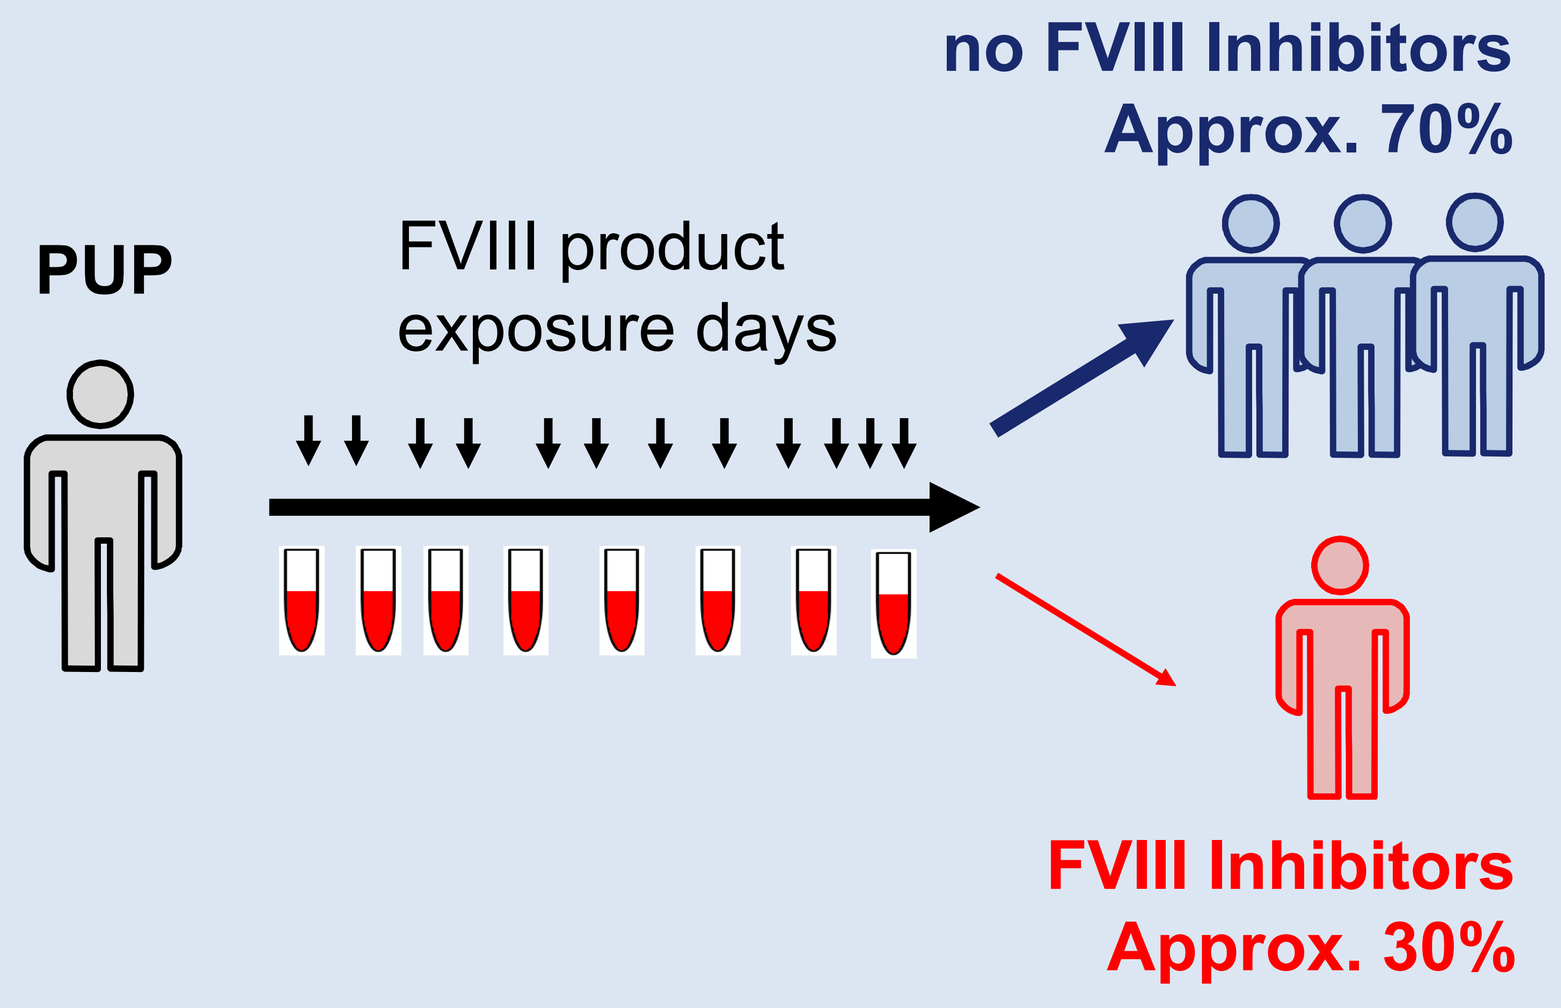
no FVIII Inhibitors
Approx. 70%
PUP
FVIII product
exposure days
FVIII Inhibitors
Approx. 30%

Figure 1 Legend: A Pre-FVIII exposure and seven post-FVIII exposure day (ED) samples (after ED 1, 5, 10, 20, 30, 40 and 50) will be available in HIPS for immune monitoring.

Table 1: HIPS laboratory assessments

Assessment	Pre-FVIII Exposure	Post-FVIII Exposure
FVIII activity	✓	
FVIII inhibitor		✓
$CD4^+$ T-cell signatures and FVIII-binding Ab	✓	✓
General immune status	✓	✓
RNA expression profile of whole blood	✓	✓
Genotyping of FVIII and other genes		✓

Results

FVIII-specific IgG1 and IgG4 were the predominant antibodies found in inhibitor patients, whereas IgG1 and IgG3 were observed in selected healthy donors and in non-inhibitor patients. Interestingly, no FVIII-specific IgG4 was detected in healthy individuals from different geographies or in patients without FVIII inhibitors (Figure 2). In addition, our data indicate an up to 100-fold higher apparent affinity of FVIII-specific antibodies found in patients with FVIII inhibitors when compared to non-inhibitor patients and healthy individuals. The highest affinity was detected for FVIII-specific IgG4 expressed by patients with FVIII inhibitors (Figure 3).

CD4+ T-cell gene expression signatures of in vitro restimulated PBMCs indicate a FVIII-specific up-regulation of pro-inflammatory genes in patients with FVIII inhibitors (Figure 4) and the absence of this pro-inflammatory signature in patients without FVIII inhibitors (Figure 5).

Figure 2: Prevalence of FVIII-specific antibodies in different cohorts

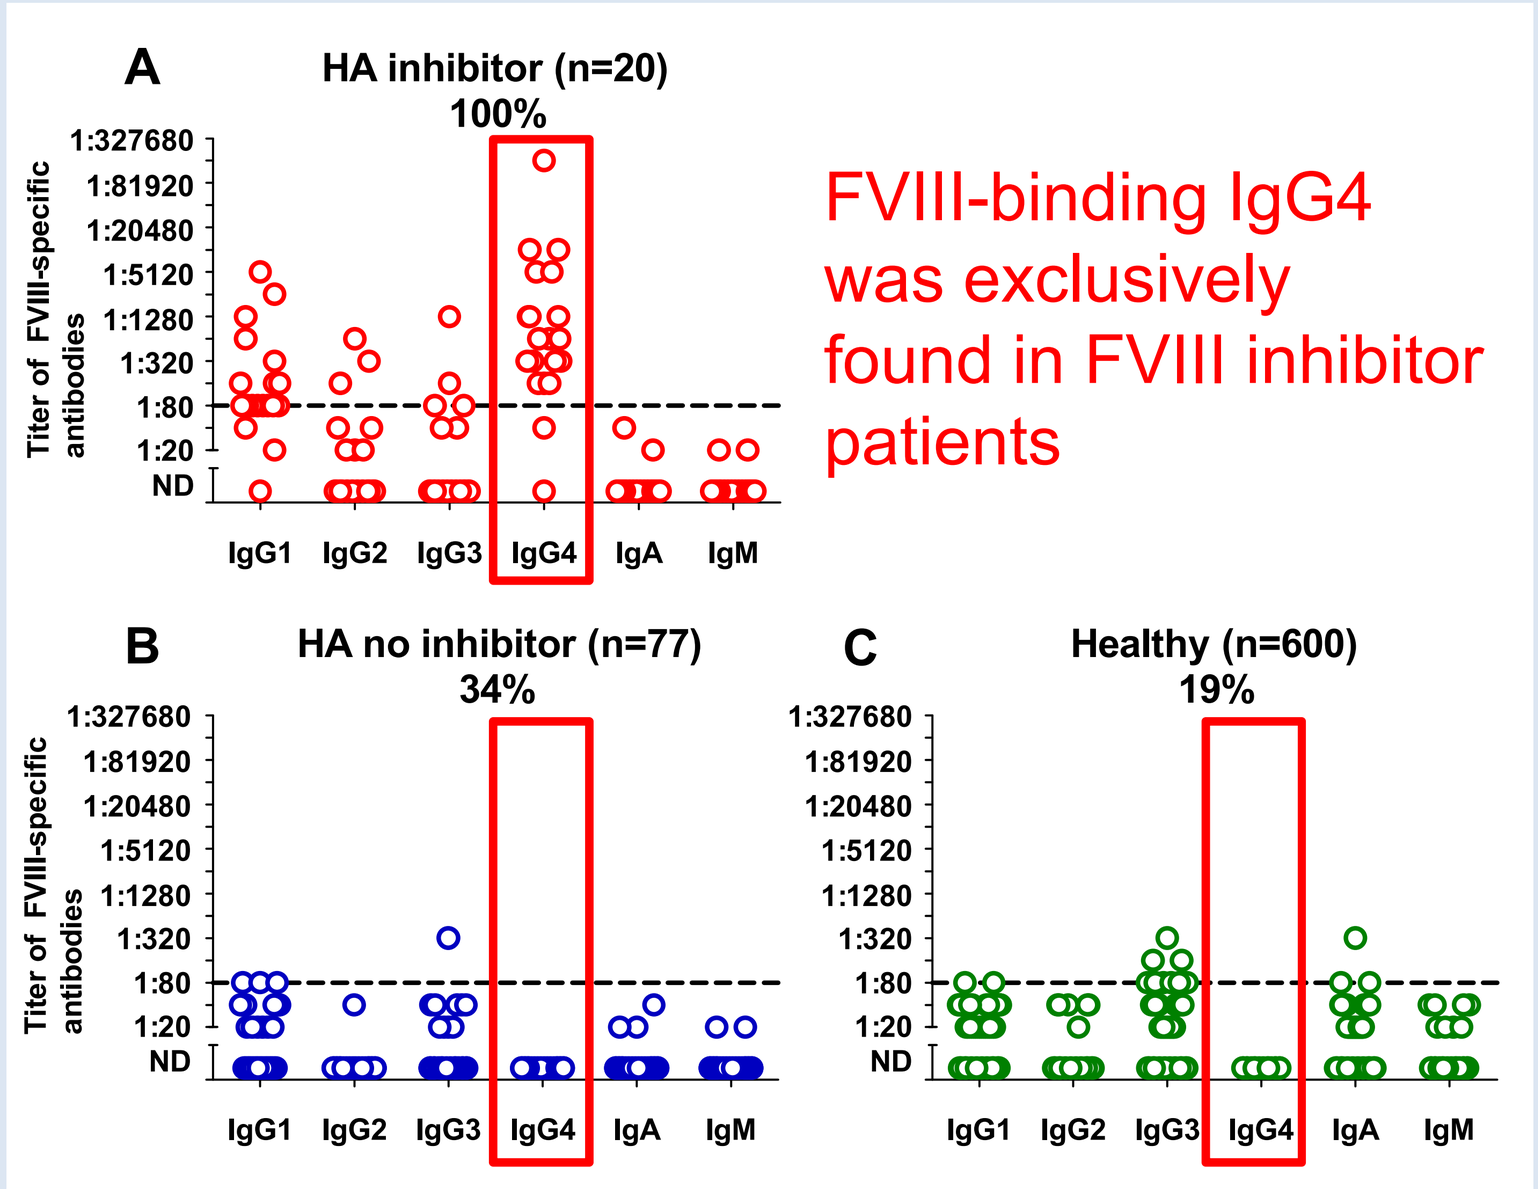
A
HA inhibitor (n=20)
100%
FVIII-binding IgG4 was exclusively found in FVIII inhibitor patients
B
HA no inhibitor (n=77)
34%
C
Healthy (n=600)
19%
Titer of FVIII-specific antibodies
IgG1 IgG2 IgG3 IgG4 IgA IgM

Figure 2 Legend: n: number of plasma samples tested, %: prevalence of FVIII-binding antibodies in specific cohort HA: severe hemophilia A, 1:20 = minimal dilution (limit of detection), 1:80 = lower limit for antibody specificity testing. Data published in Whelan et al Blood 2013 [3]

Figure 3: Affinity of FVIII-specific antibodies in different cohorts

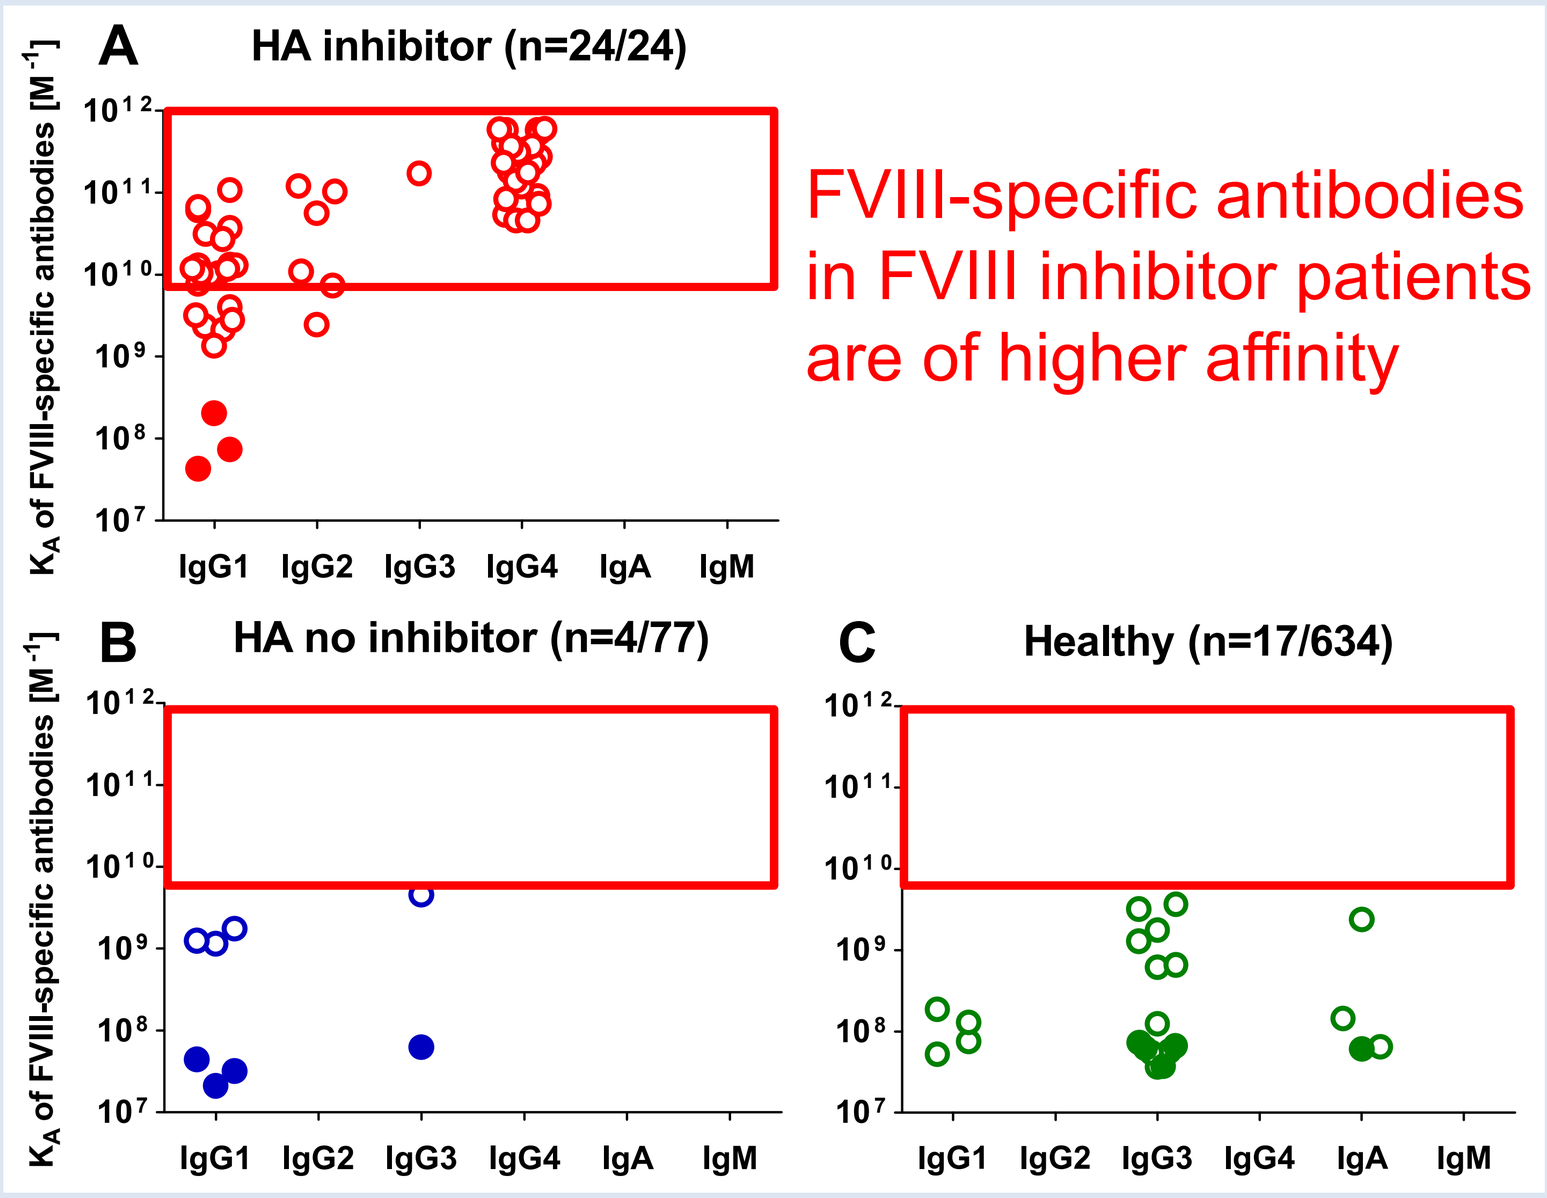
A
HA inhibitor (n=24/24)
FVIII-specific antibodies in FVIII inhibitor patients are of higher affinity
B
HA no inhibitor (n=4/77)
C
Healthy (n=17/634)
KA of FVIII-specific antibodies [M-1]
IgG1 IgG2 IgG3 IgG4 IgA IgM

Figure 3 Legend: n: number of plasma samples tested, HA: severe hemophilia A, K_A: apparent affinity, open symbols: affinity population 1, closed symbols: affinity population 2. Manuscript in preparation.

Figure 4: FVIII-specific T-cell signatures in a HA patient with FVIII inhibitor

FVIII Inhibitor [BU/ml]	Titer of FVIII-binding antibodies						Affinity of FVIII-binding antibodies [1/M]
	IgG1	IgG2	IgG3	IgG4	IgA	IgM	
1	1:40	neg	neg	1:40	neg	neg	IgG1: 1.52×10^{10} & 6.69×10^{7} IgG4: 2.22×10^{11}

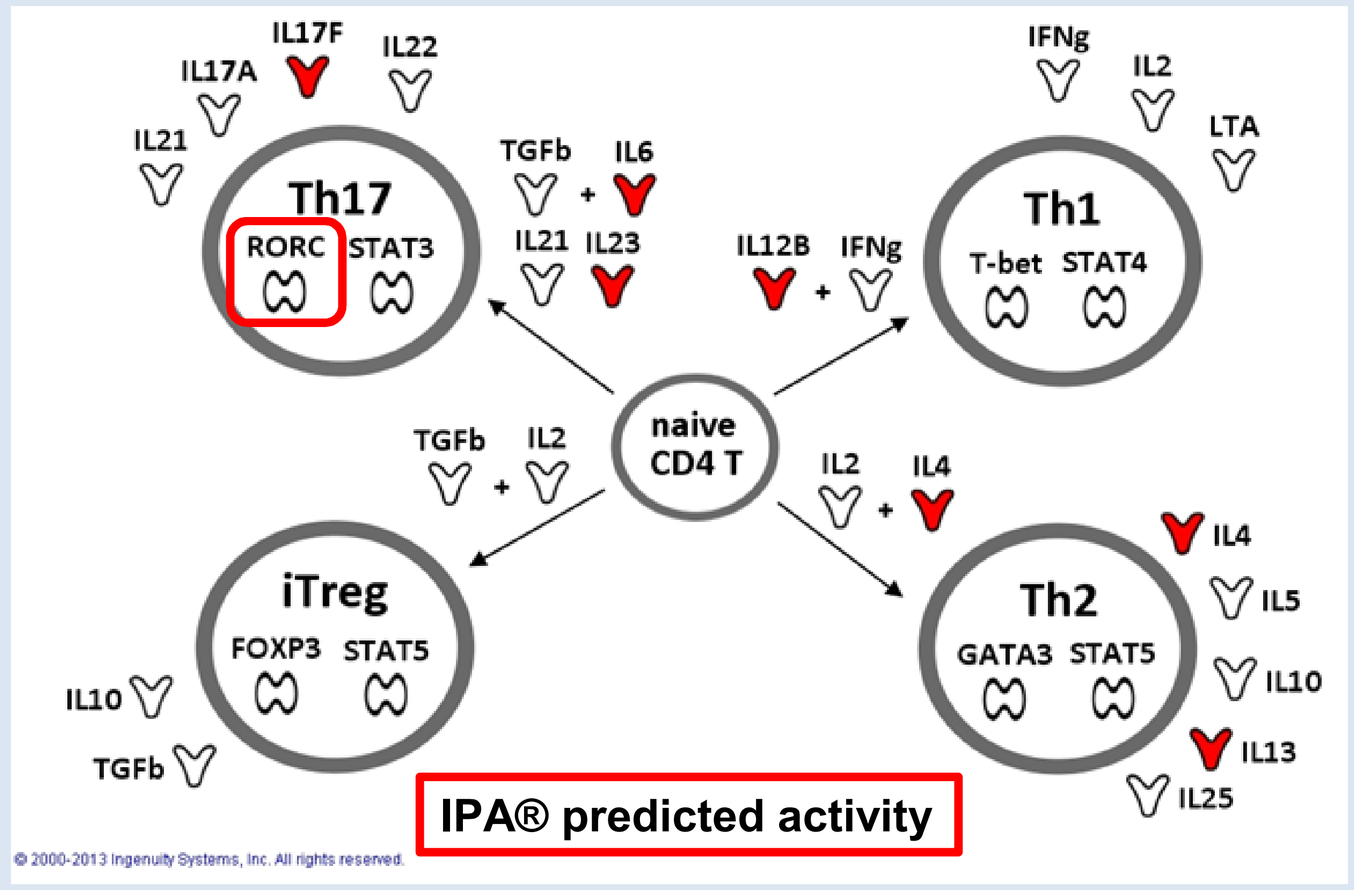
Th17
RORC STAT3
Th1
T-bet STAT4
naive CD4 T
iTreg
FOXP3 STAT5
Th2
GATA3 STAT5
IPA® predicted activity

Figure Legend: Signature transcription factors and cytokines of major CD4+ T cell subsets. RED: > 2-fold up-regulation, GREEN: > 2-fold down-regulation, WHITE: no regulation

Figure 5: FVIII-specific T-cell signatures in a HA patient without FVIII inhibitor

FVIII Inhibitor [BU/ml]	Titer of FVIII-binding antibodies						Affinity of FVIII-binding antibodies [1/M]
	IgG1	IgG2	IgG3	IgG4	IgA	IgM	
<0,4	neg	neg	neg	neg	neg	neg	not applicable

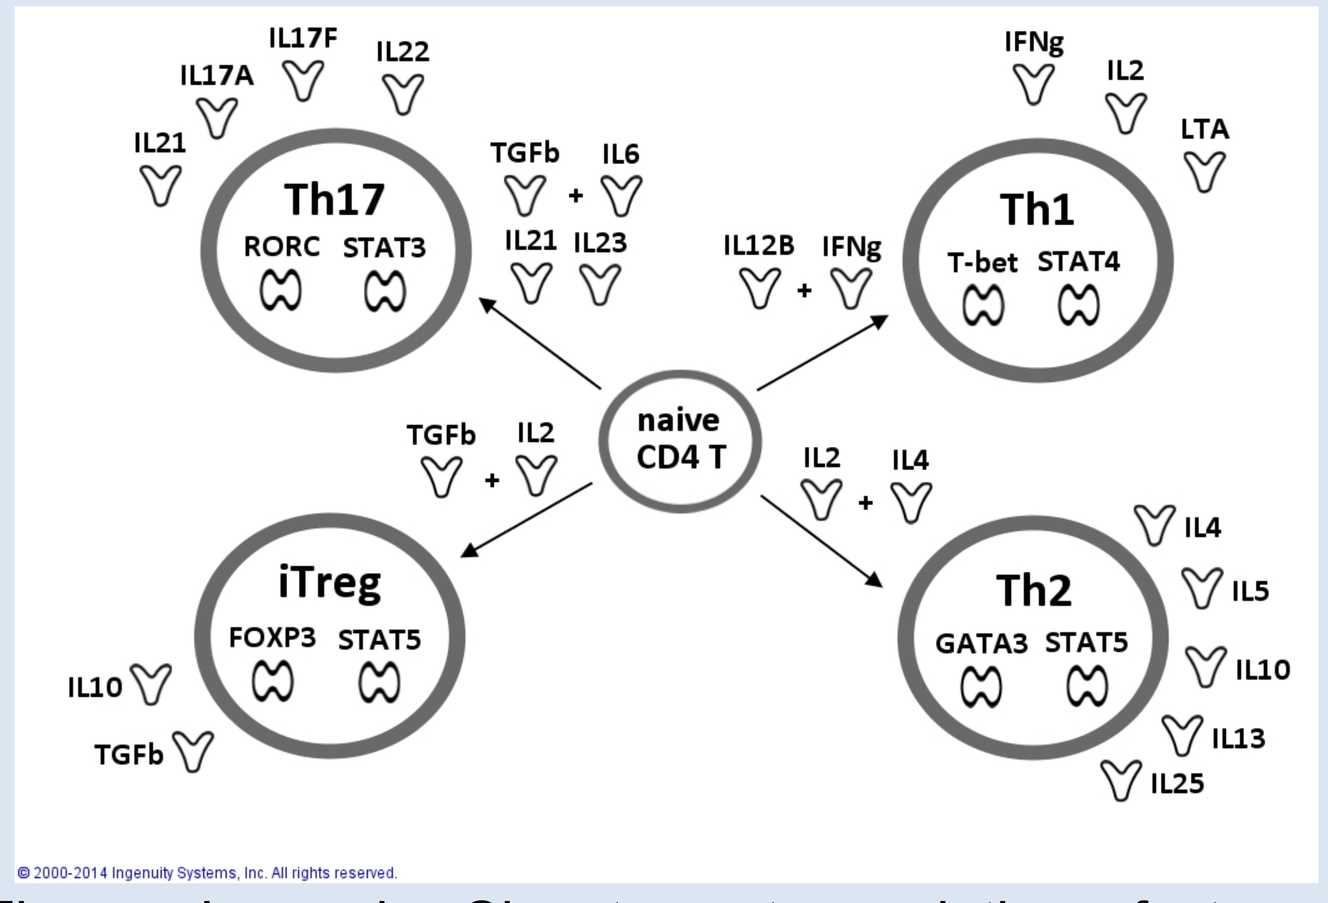
Th17
RORC STAT3
Th1
T-bet STAT4
naive CD4 T
iTreg
FOXP3 STAT5
Th2
GATA3 STAT5

Figure Legend: Signature transcription factors and cytokines of major CD4+ T cell subsets. RED: > 2-fold up-regulation, GREEN: > 2-fold down-regulation, WHITE: no regulation

Conclusions

- **Methodology to monitor FVIII-specific immune responses in 3-4ml of blood in a multi-center setting was successfully developed**
- **Application of this methodology in HIPS has the potential to provide a missing piece in understanding FVIII inhibitor development**
- **Lessons to be learned during HIPS will provide scientific basis to improve congenital hemophilia A care**

References

1. Ragni MV et al (1989) Disappearance of inhibitor to factor III in HIV-infected hemophiliacs with progression to AIDS or severe ARC. Transfusion, 29, 447-449
2. Steinitz KN et al (2012) CD4+ T-cell epitopes associated with antibody responses after intravenously and subcutaneously applied human FVIII in humanized hemophilic E17 HLA-DRB1*1501 mice. Blood, 119, 4073-4082
3. Whelan SFJ et al (2013) Distinct characteristics of antibody responses against factor VIII in healthy individuals and in different cohorts of hemophilia A patients. Blood, 121, 1039-1048
4. Bobrovnik et al. (2010) Traditional ELISA methods for antibody affinity determination fail to reveal the presence of low affinity antibodies : an alternative approach. J Mol Recognit, 23, 448-456

Presented at the WFH 2014 World Congress in Melbourne, Australia • May 11-15, 2014

If you have any additional questions, please feel free to contact Baxter Bioscience Medical Information at medinfo@baxter.com.

Conflicts of interest: Hofbauer CJ, Scheiflinger F and Reipert BM are employees of Baxter Bioscience

WFH2014 44-P-T

Inhibitors, Pathogenesis, Prevention and Treatment

Christoph Hofbauer

DOI: 10.3252/pso.eu.WFH2014.2014

Poster presented at: