The UK Buddy Award Swimming Academy

P-W-201

Bladen M1, McLaughlin P2, Spires J1, Bridge J3, Scanlon A4, Gibson J5, Hart DP6.

Category: Physiotherapy and Rehabilitation

Introduction & Objectives

Swimming is a popular activity for children in the UK. However, even with the skill of being able to swim, only a minority continue to swim regularly through adolescence and into adulthood. Swimming is an acceptable activity for individuals living with haemophilia regardless of joint status and offers a marvelous opportunity for cardiovascular and musculoskeletal health. To maximise the value of swimming, boys need to engage with swimming technically and require inspiration and support to help establish it as part of their lives.

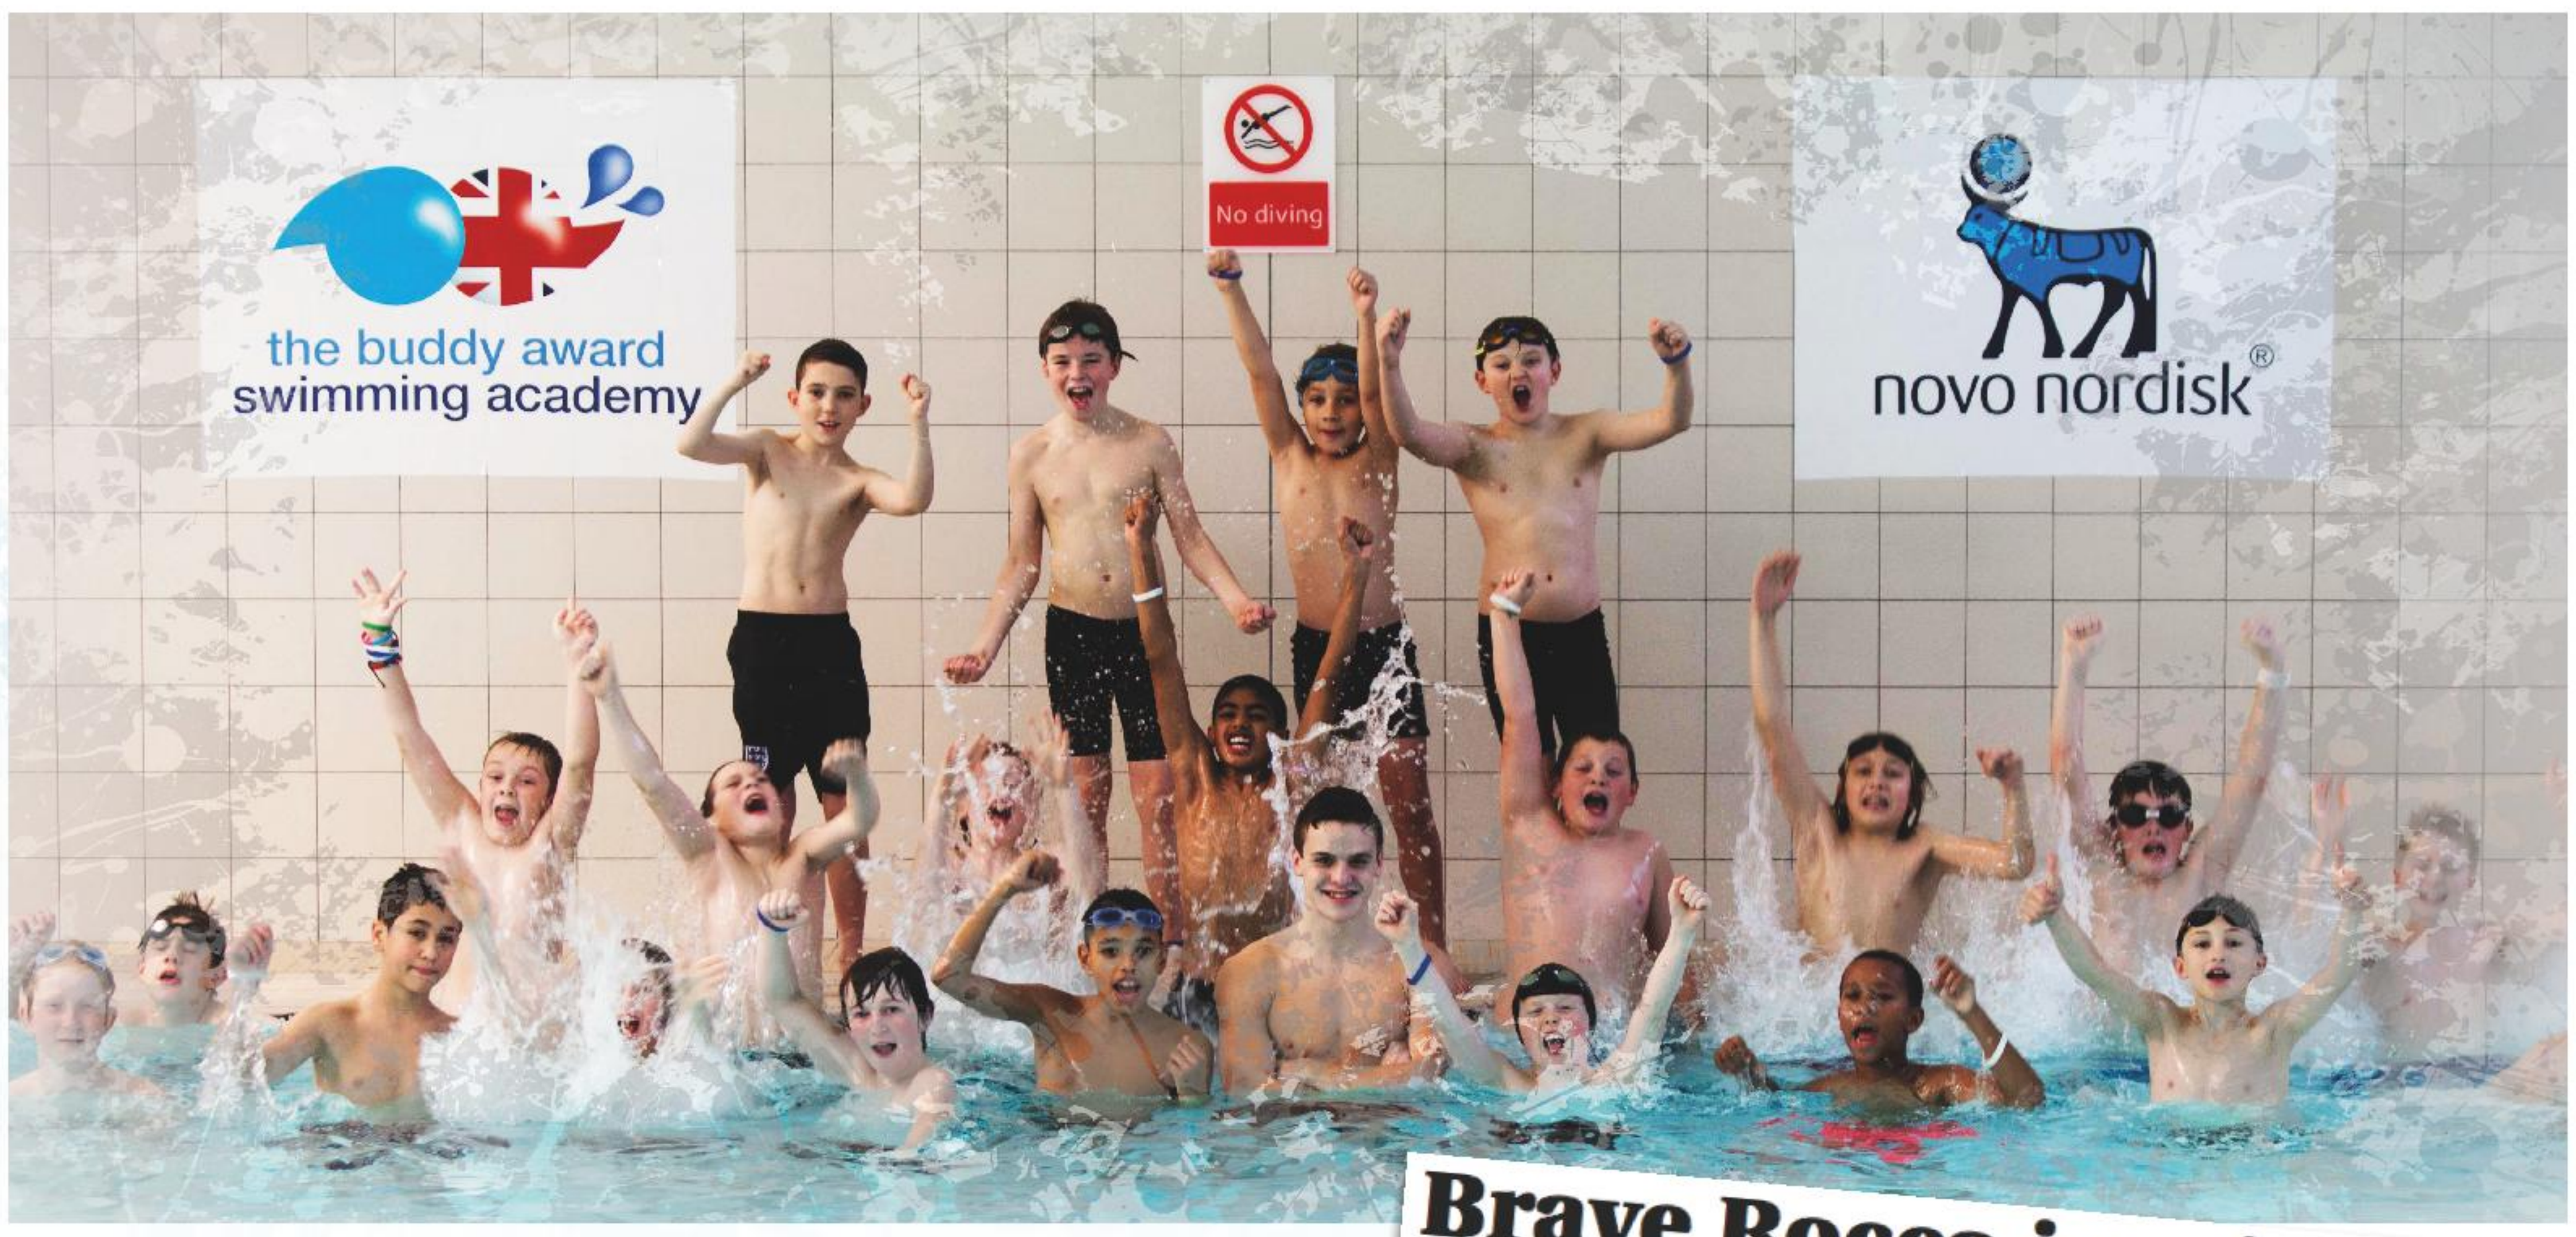
the buddy award
swimming academy
No diving
novo nordisk

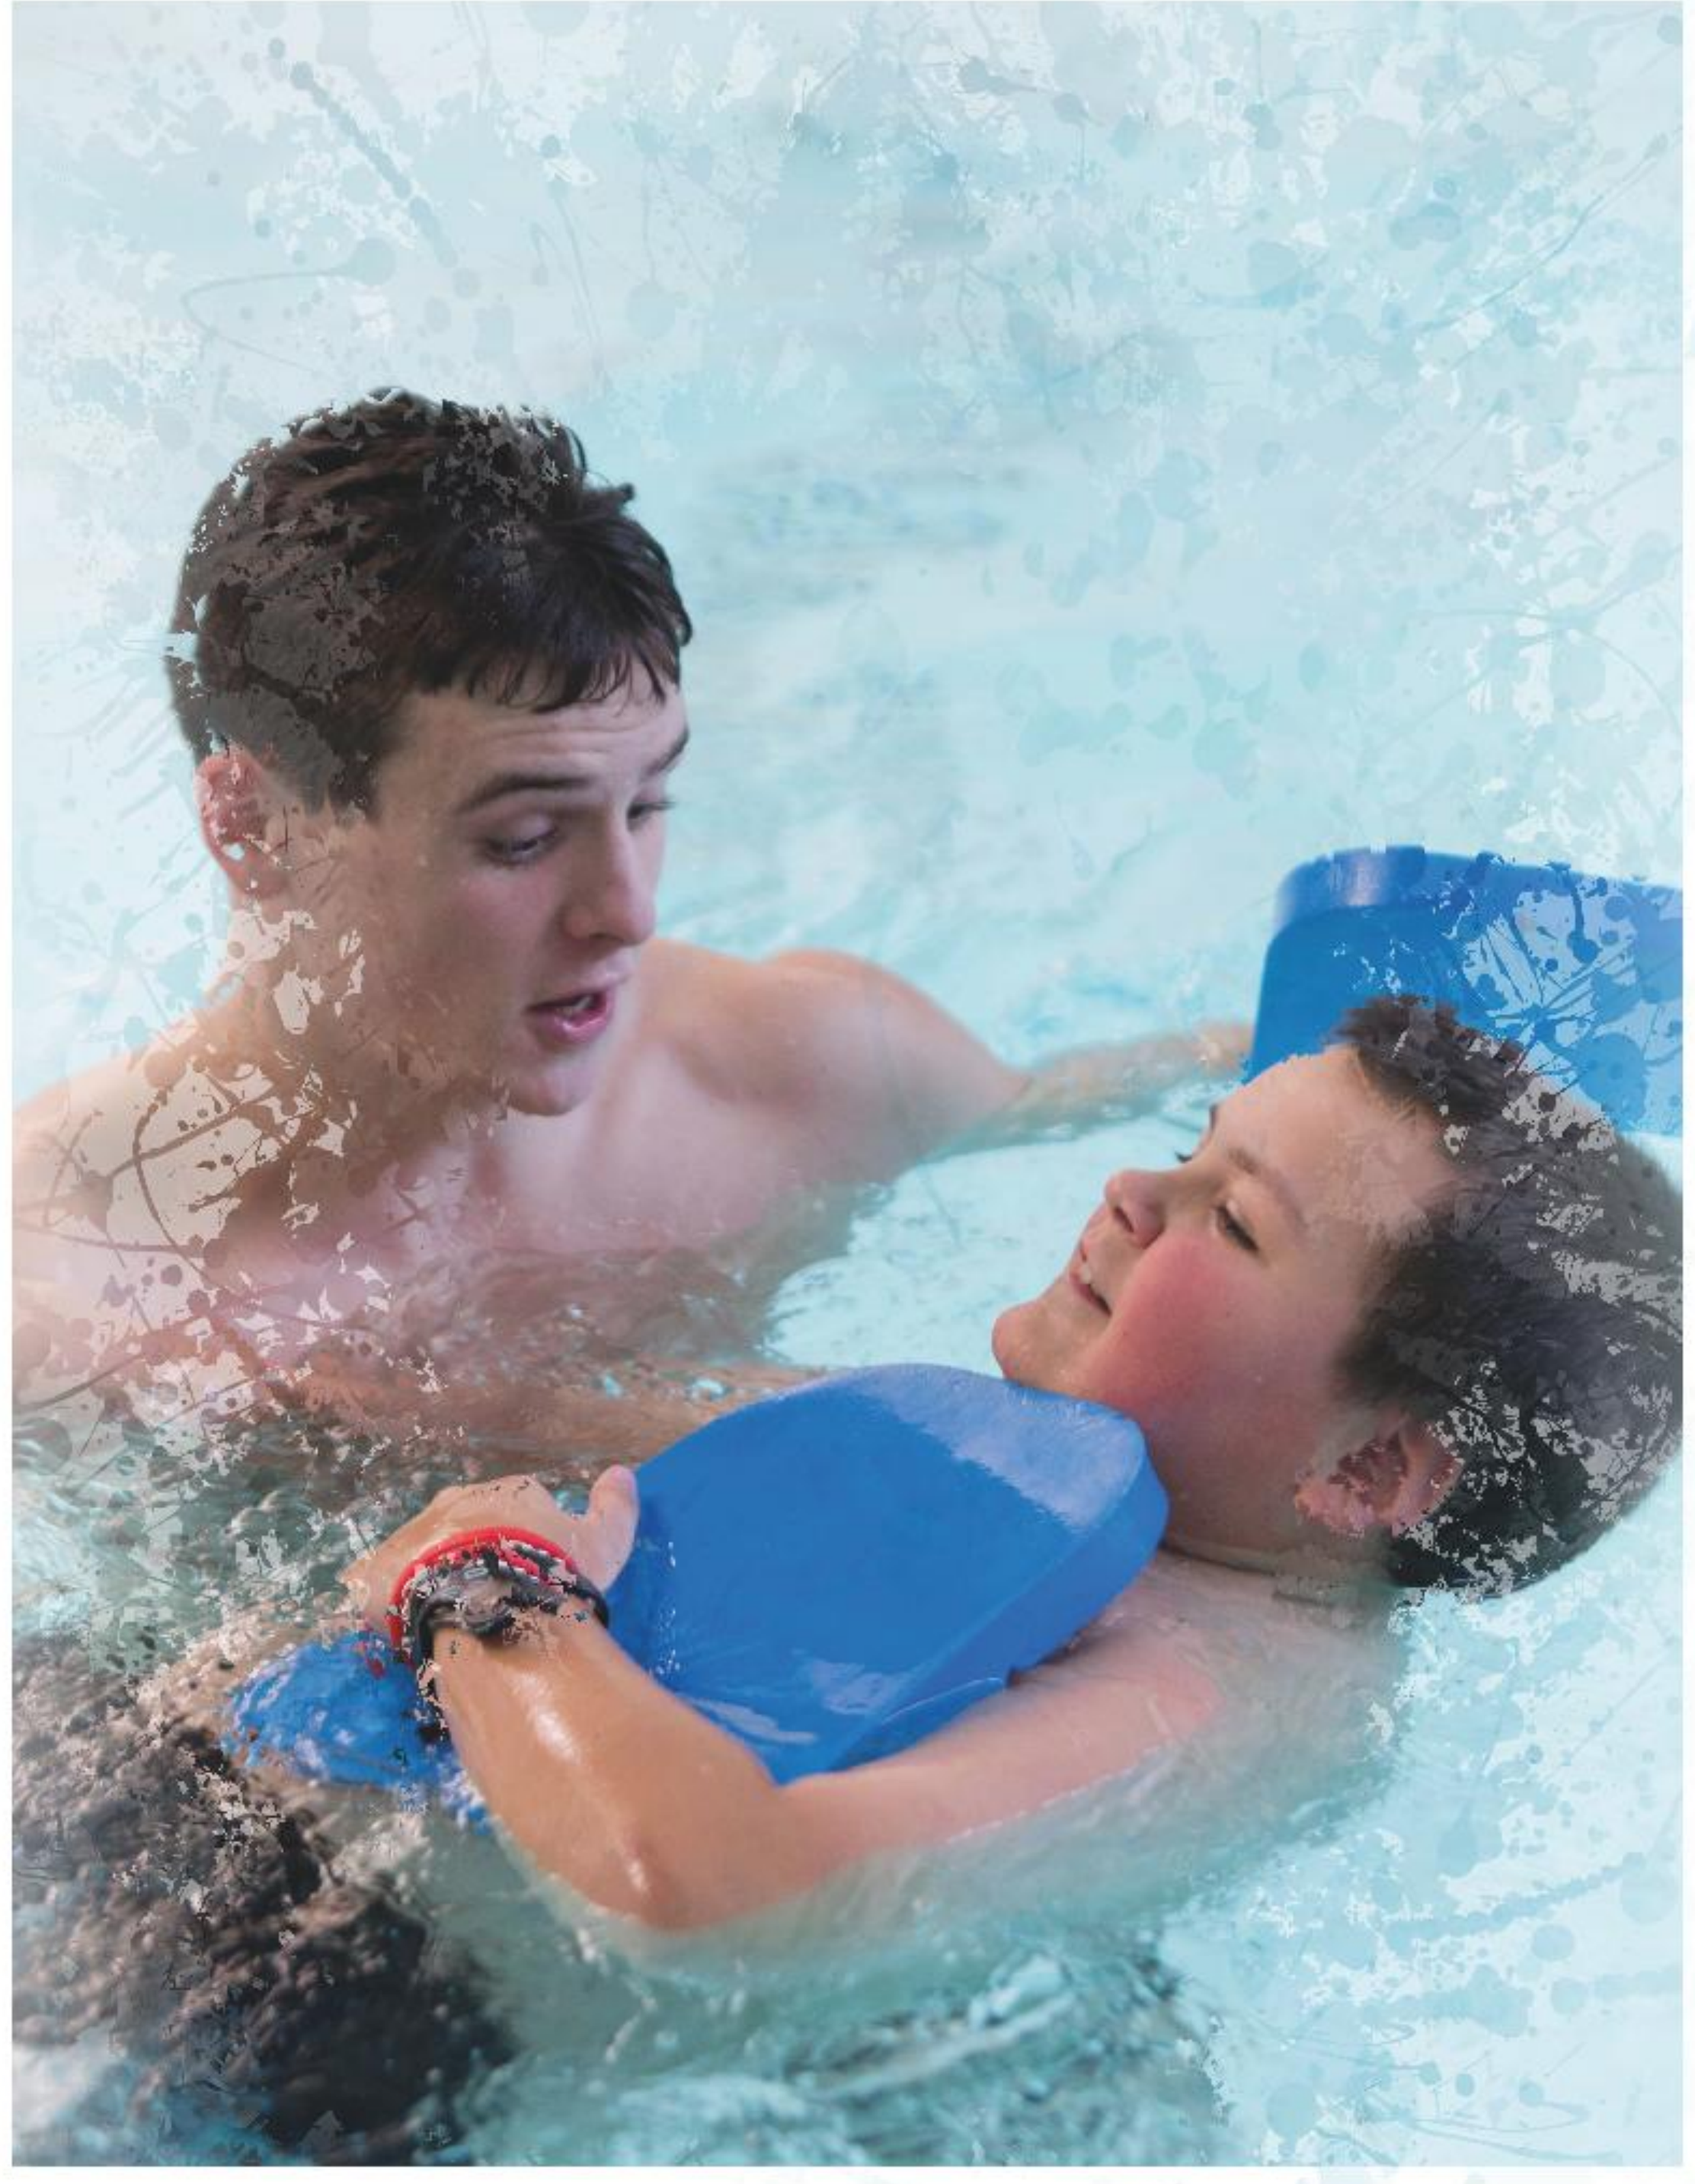

Method

The Buddy Award Swimming Academy, sponsored by Novo Nordisk UK, was developed by an eight-member steering committee, consisting of leading doctors, nurses, physiotherapists and swimming coaches. The academy aimed to inspire boys living with haemophilia to improve their swimming technique and see it as a fun healthy activity. The inaugural group training session provided haemophilia specific and swimming specific training. Jack Bridge, a young Paralympic (London 2012) breast stroke specialist, aged 19, who has severe haemophilia A and a history of an inhibitor, provided the key inspirational link between haemophilia and swimming excellence. Jack and two Team GB swimming coaches, also part of the steering committee, helped to design and deliver the swimming programme.

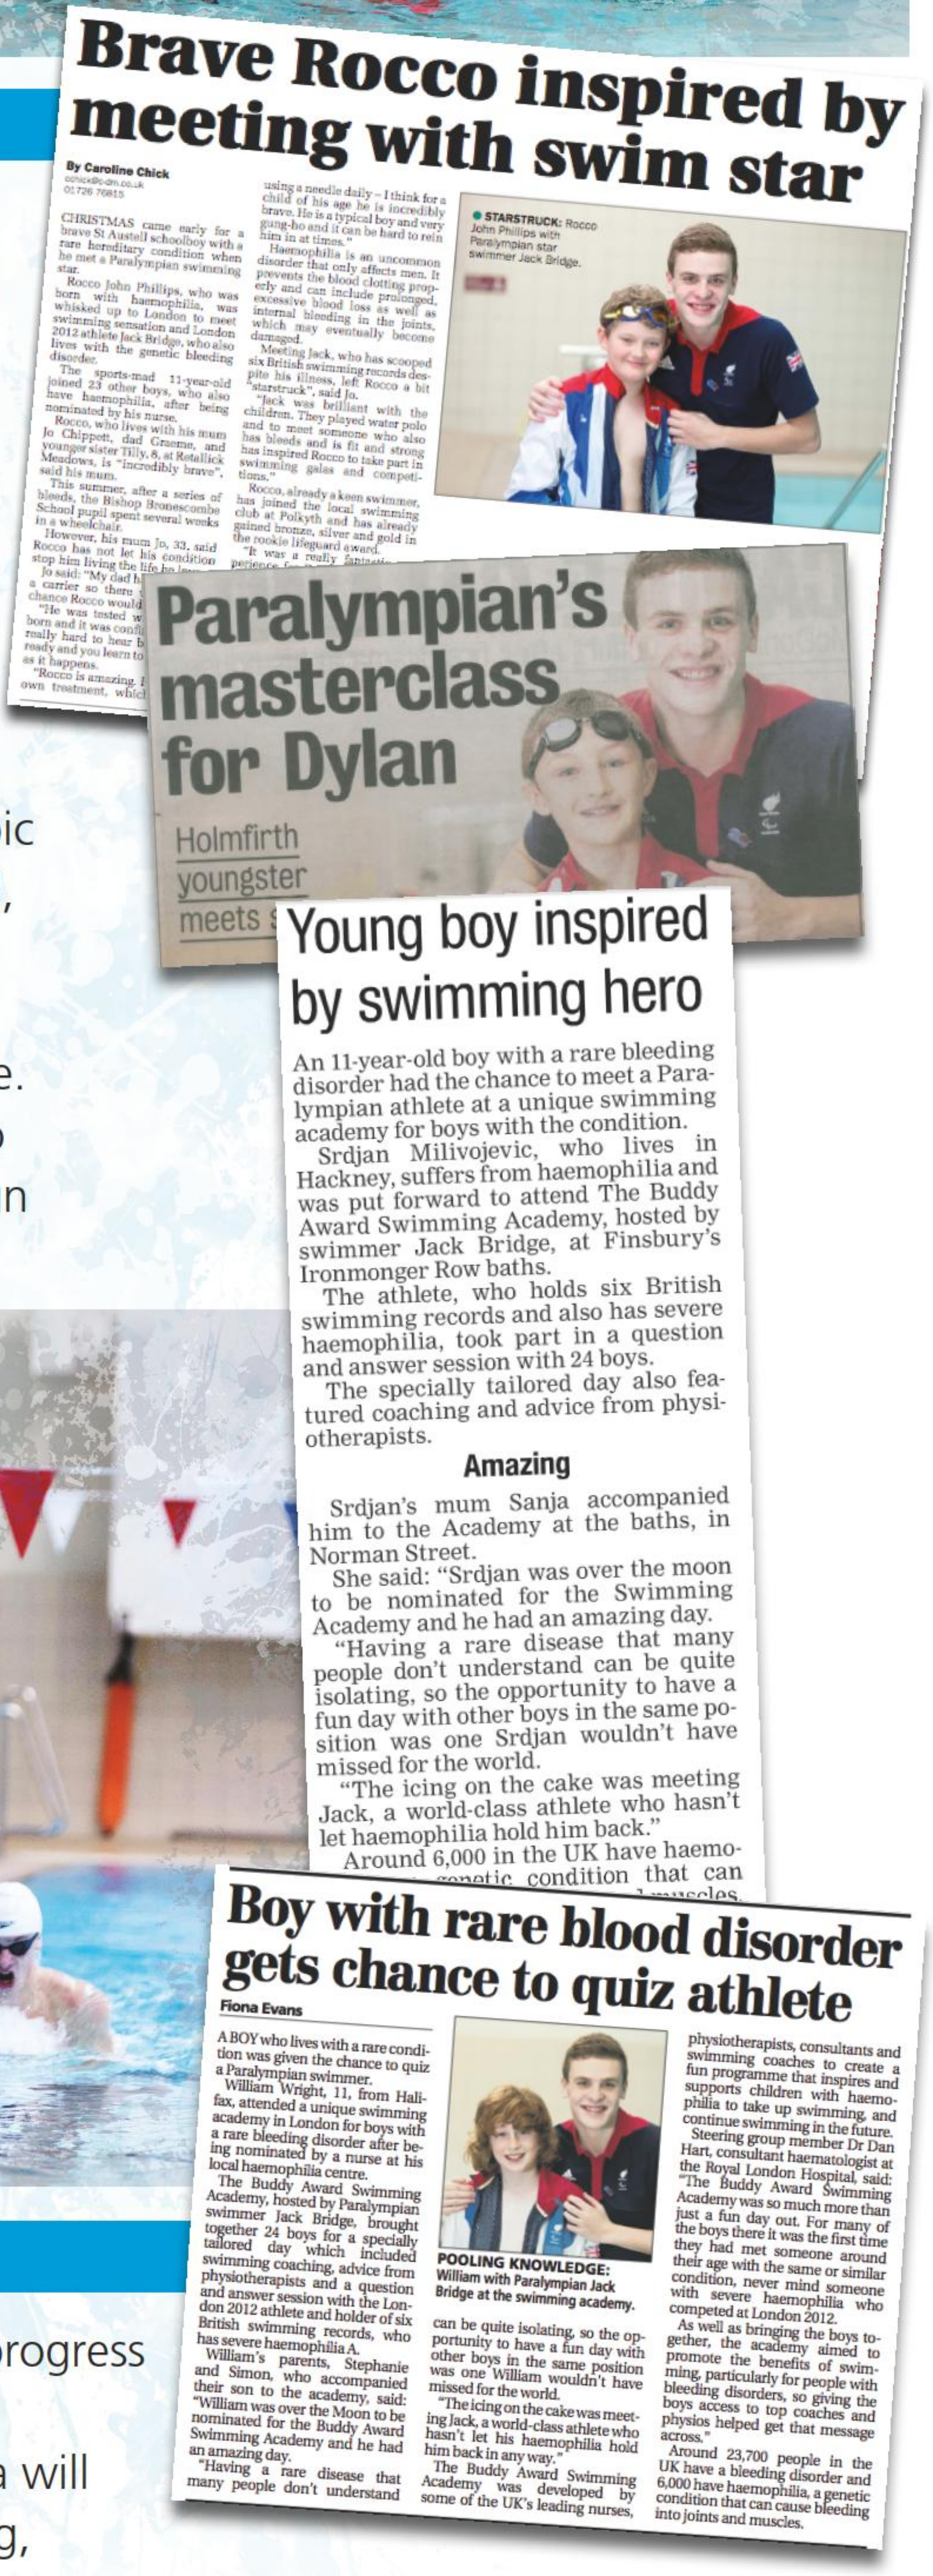

Brave Rocco inspired by meeting with swim star

By Caroline Chick
cchick@c-dm.co.uk
01726 76815

CHRISTMAS came early for a brave St Austell schoolboy with a rare hereditary condition when he met a Paralympian swimming star.

Rocco John Phillips, who was born with haemophilia, was whisked up to London to meet swimming sensation and London 2012 athlete Jack Bridge, who also lives with the genetic bleeding disorder.

The sports-mad 11-year-old joined 23 other boys, who also have haemophilia, after being nominated by his nurse.

Rocco, who lives with his mum Jo Chippett, dad Graeme, and younger sister Tilly, 8, at Retallick Meadows, is "incredibly brave", said his mum.

This summer, after a series of bleeds, the Bishop Bronescombe School pupil spent several weeks in a wheelchair.

However, his mum Jo, 33, said Rocco has not let his condition stop him living the life he lo...

using a needle daily – I think for a child of his age he is incredibly brave. He is a typical boy and very gung-ho and it can be hard to rein him in at times."

Haemophilia is an uncommon disorder that only affects men. It prevents the blood clotting properly and can include prolonged, excessive blood loss as well as internal bleeding in the joints, which may eventually become damaged.

Meeting Jack, who has scooped six British swimming records despite his illness, left Rocco a bit "starstruck", said Jo.

"Jack was brilliant with the children. They played water polo and to meet someone who also has bleeds and is fit and strong has inspired Rocco to take part in swimming galas and competitions."

Rocco, already a keen swimmer, has joined the local swimming club at Polkyth and has already gained bronze, silver and gold in the rookie lifeguard award.

STARSTRUCK: Rocco John Phillips with Paralympian star swimmer Jack Bridge.

Paralympian's masterclass for Dylan

Holmfirth youngster meets

Young boy inspired by swimming hero

An 11-year-old boy with a rare bleeding disorder had the chance to meet a Paralympian athlete at a unique swimming academy for boys with the condition.

Srdjan Milivojevic, who lives in Hackney, suffers from haemophilia and was put forward to attend The Buddy Award Swimming Academy, hosted by swimmer Jack Bridge, at Finsbury's Ironmonger Row baths.

The athlete, who holds six British swimming records and also has severe haemophilia, took part in a question and answer session with 24 boys.

The specially tailored day also featured coaching and advice from physiotherapists.

Amazing

Srdjan's mum Sanja accompanied him to the Academy at the baths, in Norman Street.

She said: "Srdjan was over the moon to be nominated for the Swimming Academy and he had an amazing day.

"Having a rare disease that many people don't understand can be quite isolating, so the opportunity to have a fun day with other boys in the same position was one Srdjan wouldn't have missed for the world.

"The icing on the cake was meeting Jack, a world-class athlete who hasn't let haemophilia hold him back."

Around 6,000 in the UK have haemo-

Boy with rare blood disorder gets chance to quiz athlete

Fiona Evans

A BOY who lives with a rare condition was given the chance to quiz a Paralympian swimmer.

William Wright, 11, from Halifax, attended a unique swimming academy in London for boys with a rare bleeding disorder after being nominated by a nurse at his local haemophilia centre.

The Buddy Award Swimming Academy, hosted by Paralympian swimmer Jack Bridge, brought together 24 boys for a specially tailored day which included swimming coaching, advice from physiotherapists and a question and answer session with the London 2012 athlete and holder of six British swimming records, who has severe haemophilia A.

William's parents, Stephanie and Simon, who accompanied their son to the academy, said: "William was over the Moon to be nominated for the Buddy Award Swimming Academy and he had an amazing day.

"Having a rare disease that many people don't understand can be quite isolating, so the opportunity to have a fun day with other boys in the same position was one William wouldn't have missed for the world.

"The icing on the cake was meeting Jack, a world-class athlete who hasn't let his haemophilia hold him back in any way."

The Buddy Award Swimming Academy was developed by some of the UK's leading nurses, physiotherapists, consultants and swimming coaches to create a fun programme that inspires and supports children with haemophilia to take up swimming, and continue swimming in the future.

Steering group member Dr Dan Hart, consultant haematologist at the Royal London Hospital, said: "The Buddy Award Swimming Academy was so much more than just a fun day out. For many of the boys there it was the first time they had met someone around their age with the same or similar condition, never mind someone with severe haemophilia who competed at London 2012.

As well as bringing the boys together, the academy aimed to promote the benefits of swimming, particularly for people with bleeding disorders, so giving the boys access to top coaches and physios helped get that message across."

Around 23,700 people in the UK have a bleeding disorder and 6,000 have haemophilia, a genetic condition that can cause bleeding into joints and muscles.

POOLING KNOWLEDGE: William with Paralympian Jack Bridge at the swimming academy.

Results

Twenty-four boys (aged 9-13years) from 16 UK haemophilia centres attended the inaugural event in November 2013. Of the participants 13 described their swimming capability as "strong", 7 as "intermediate", 4 as "beginner" prior to the event. **Before they attended the Academy, 23 boys had never met any other boys with haemophilia and only one boy had other 'buddies'.** A media campaign was developed, resulting in a range of print and online coverage, with a combined circulation of 165,000.

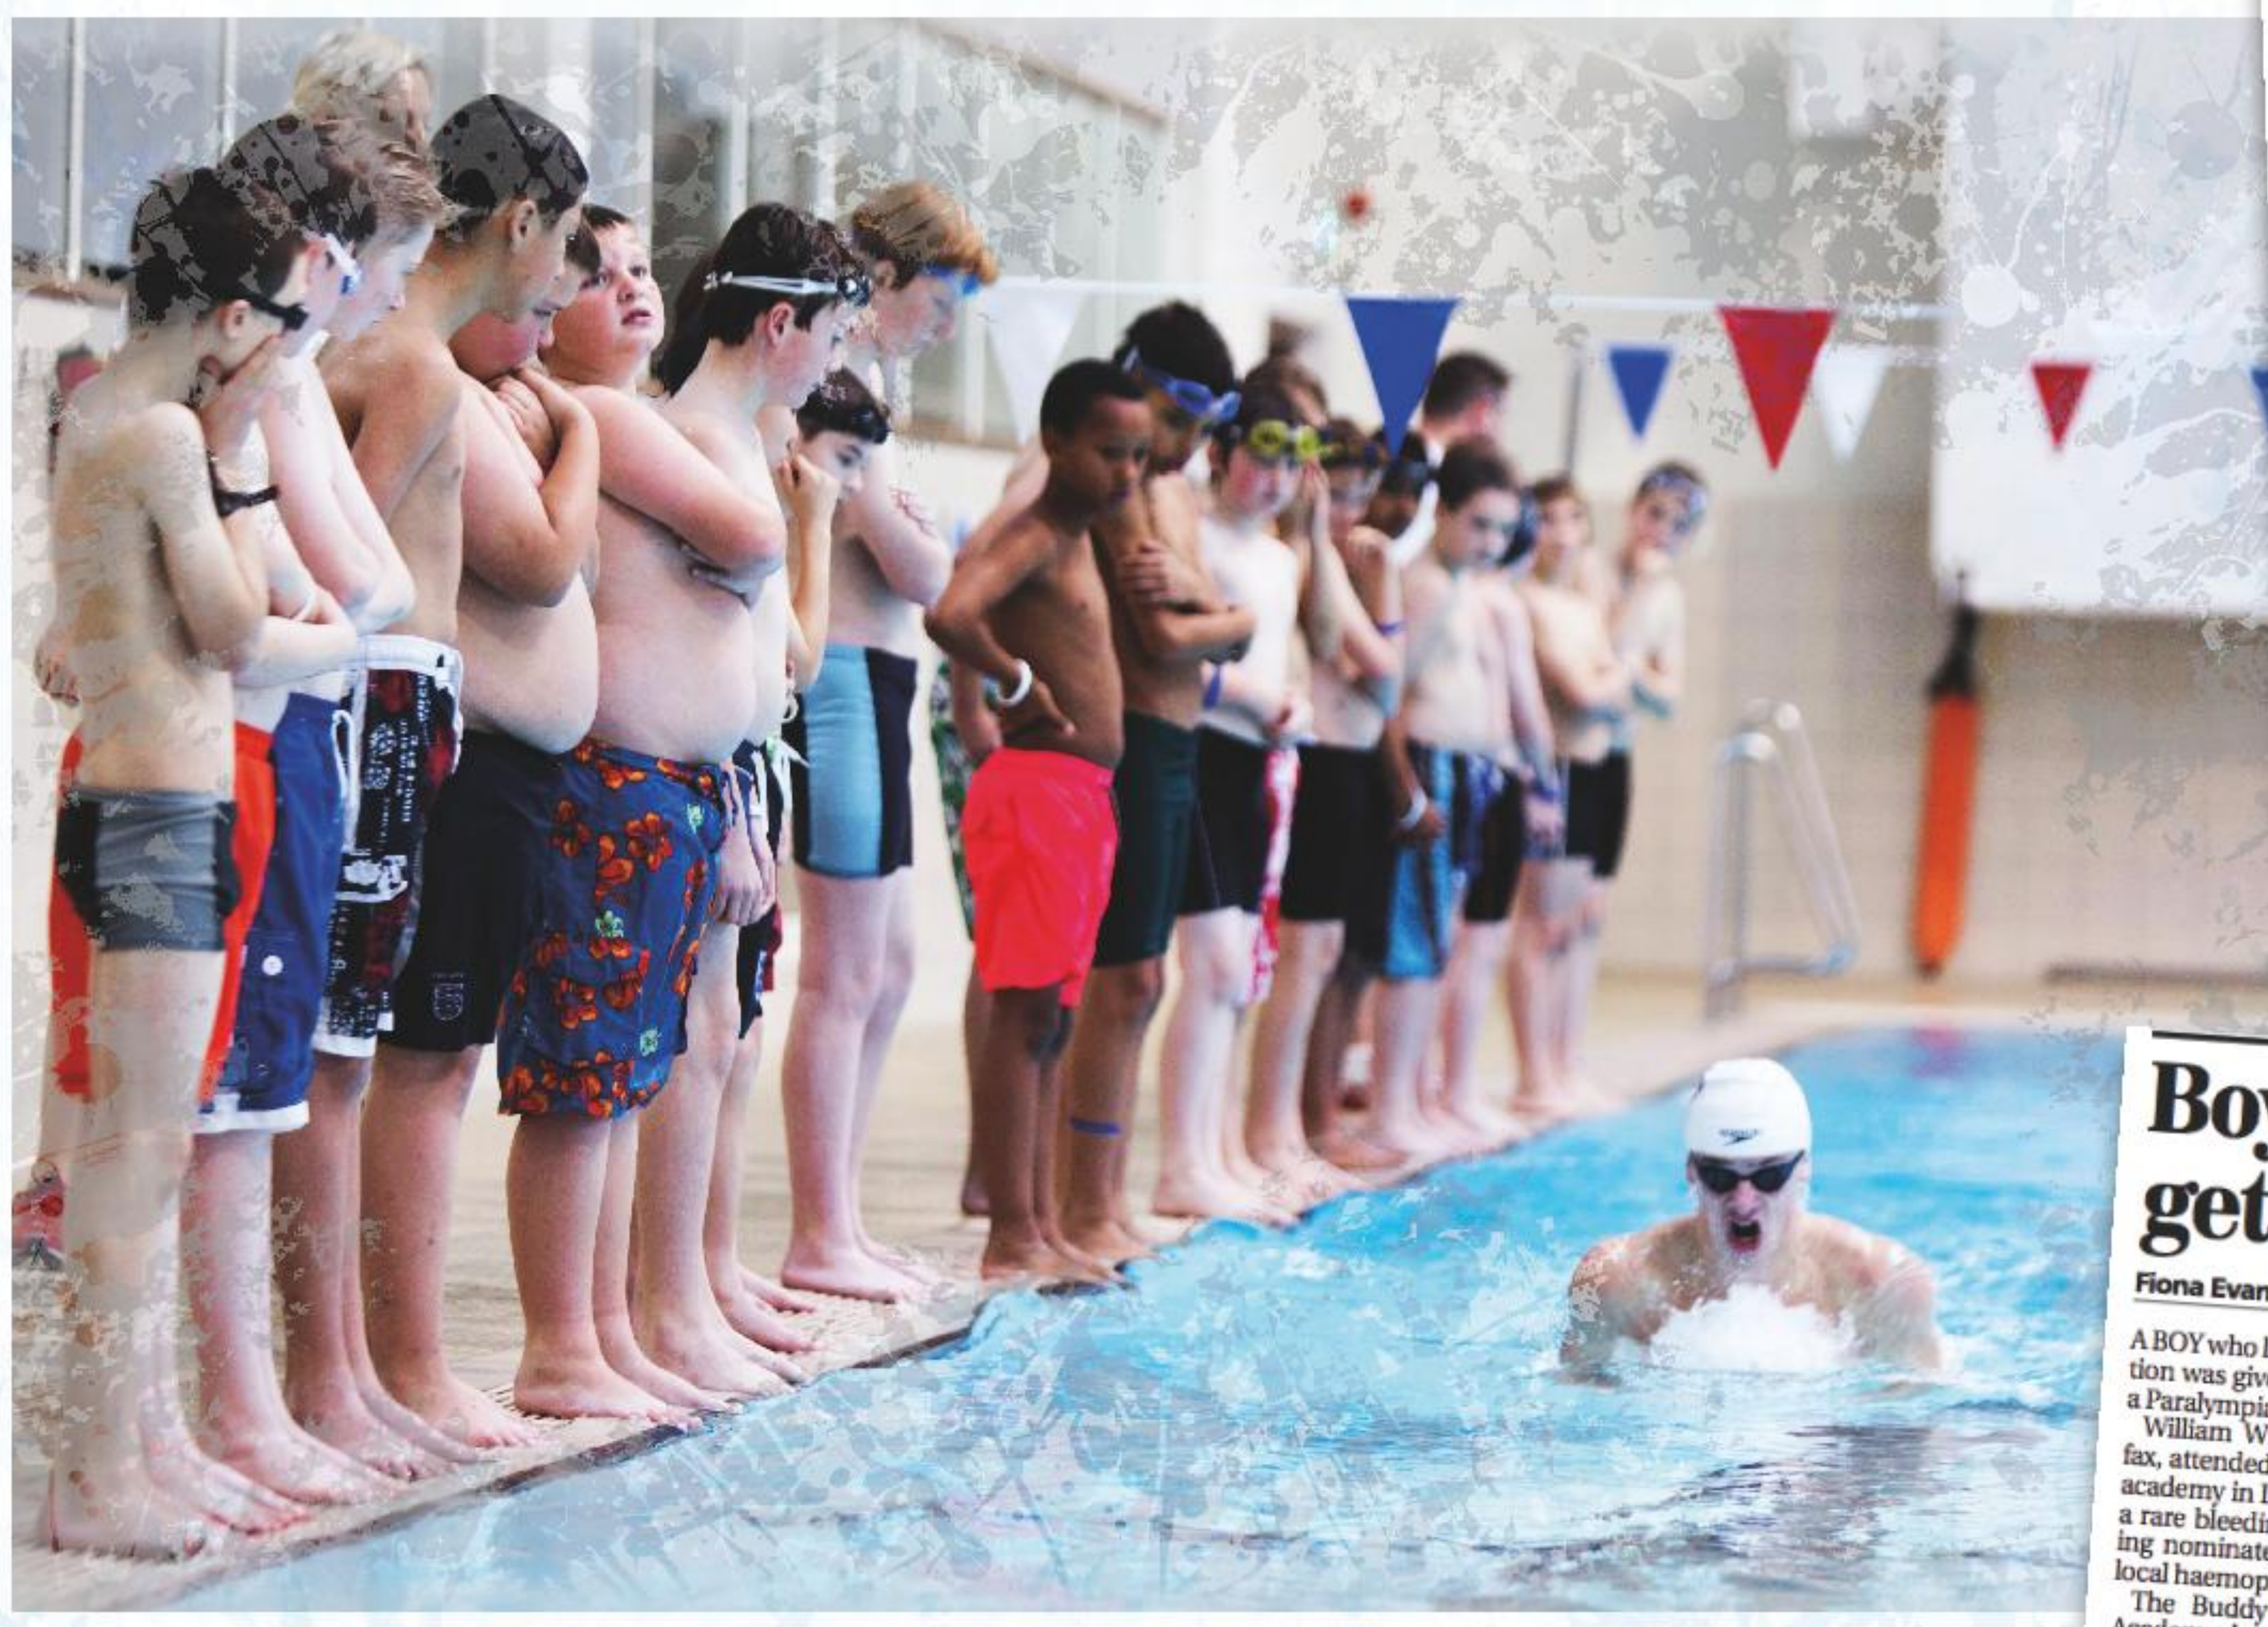

Authors:

1. Great Ormond Street Hospital, London
2. Royal Free Hospital, London
3. Paralympics GB Athlete
4. Performance and Development Coach
5. Novo Nordisk UK
6. The Royal London Hospital

Date of Preparation: March 2014; UK/CPIH/0314/0005

Conclusion

A programme roll out is planned to follow up progress and engage new recruits. It is hoped a national academy event for boys living with haemophilia will provide an attractive focus for on-going training, improvement and contact with buddies.

The Buddy Award Swimming Academy was funded by Novo Nordisk Ltd.

changing possibilities in haemophilia®

novo nordisk®

INT-WFH-Abstract-Poster-A-4.indd 1
09/04/2014 10:21

WFH2014
201--P-W
Physiotherapy and Rehabilitation
Daniel Hart
DOI: 10.3252/pso.eu.WFH2014.2014
Poster presented at: WFH 2014 World Congress Melbourne, Australia · May 11-15
PosterSessionOnline.com