Men Helping Women

An Empowering Approach to create Women Leaders

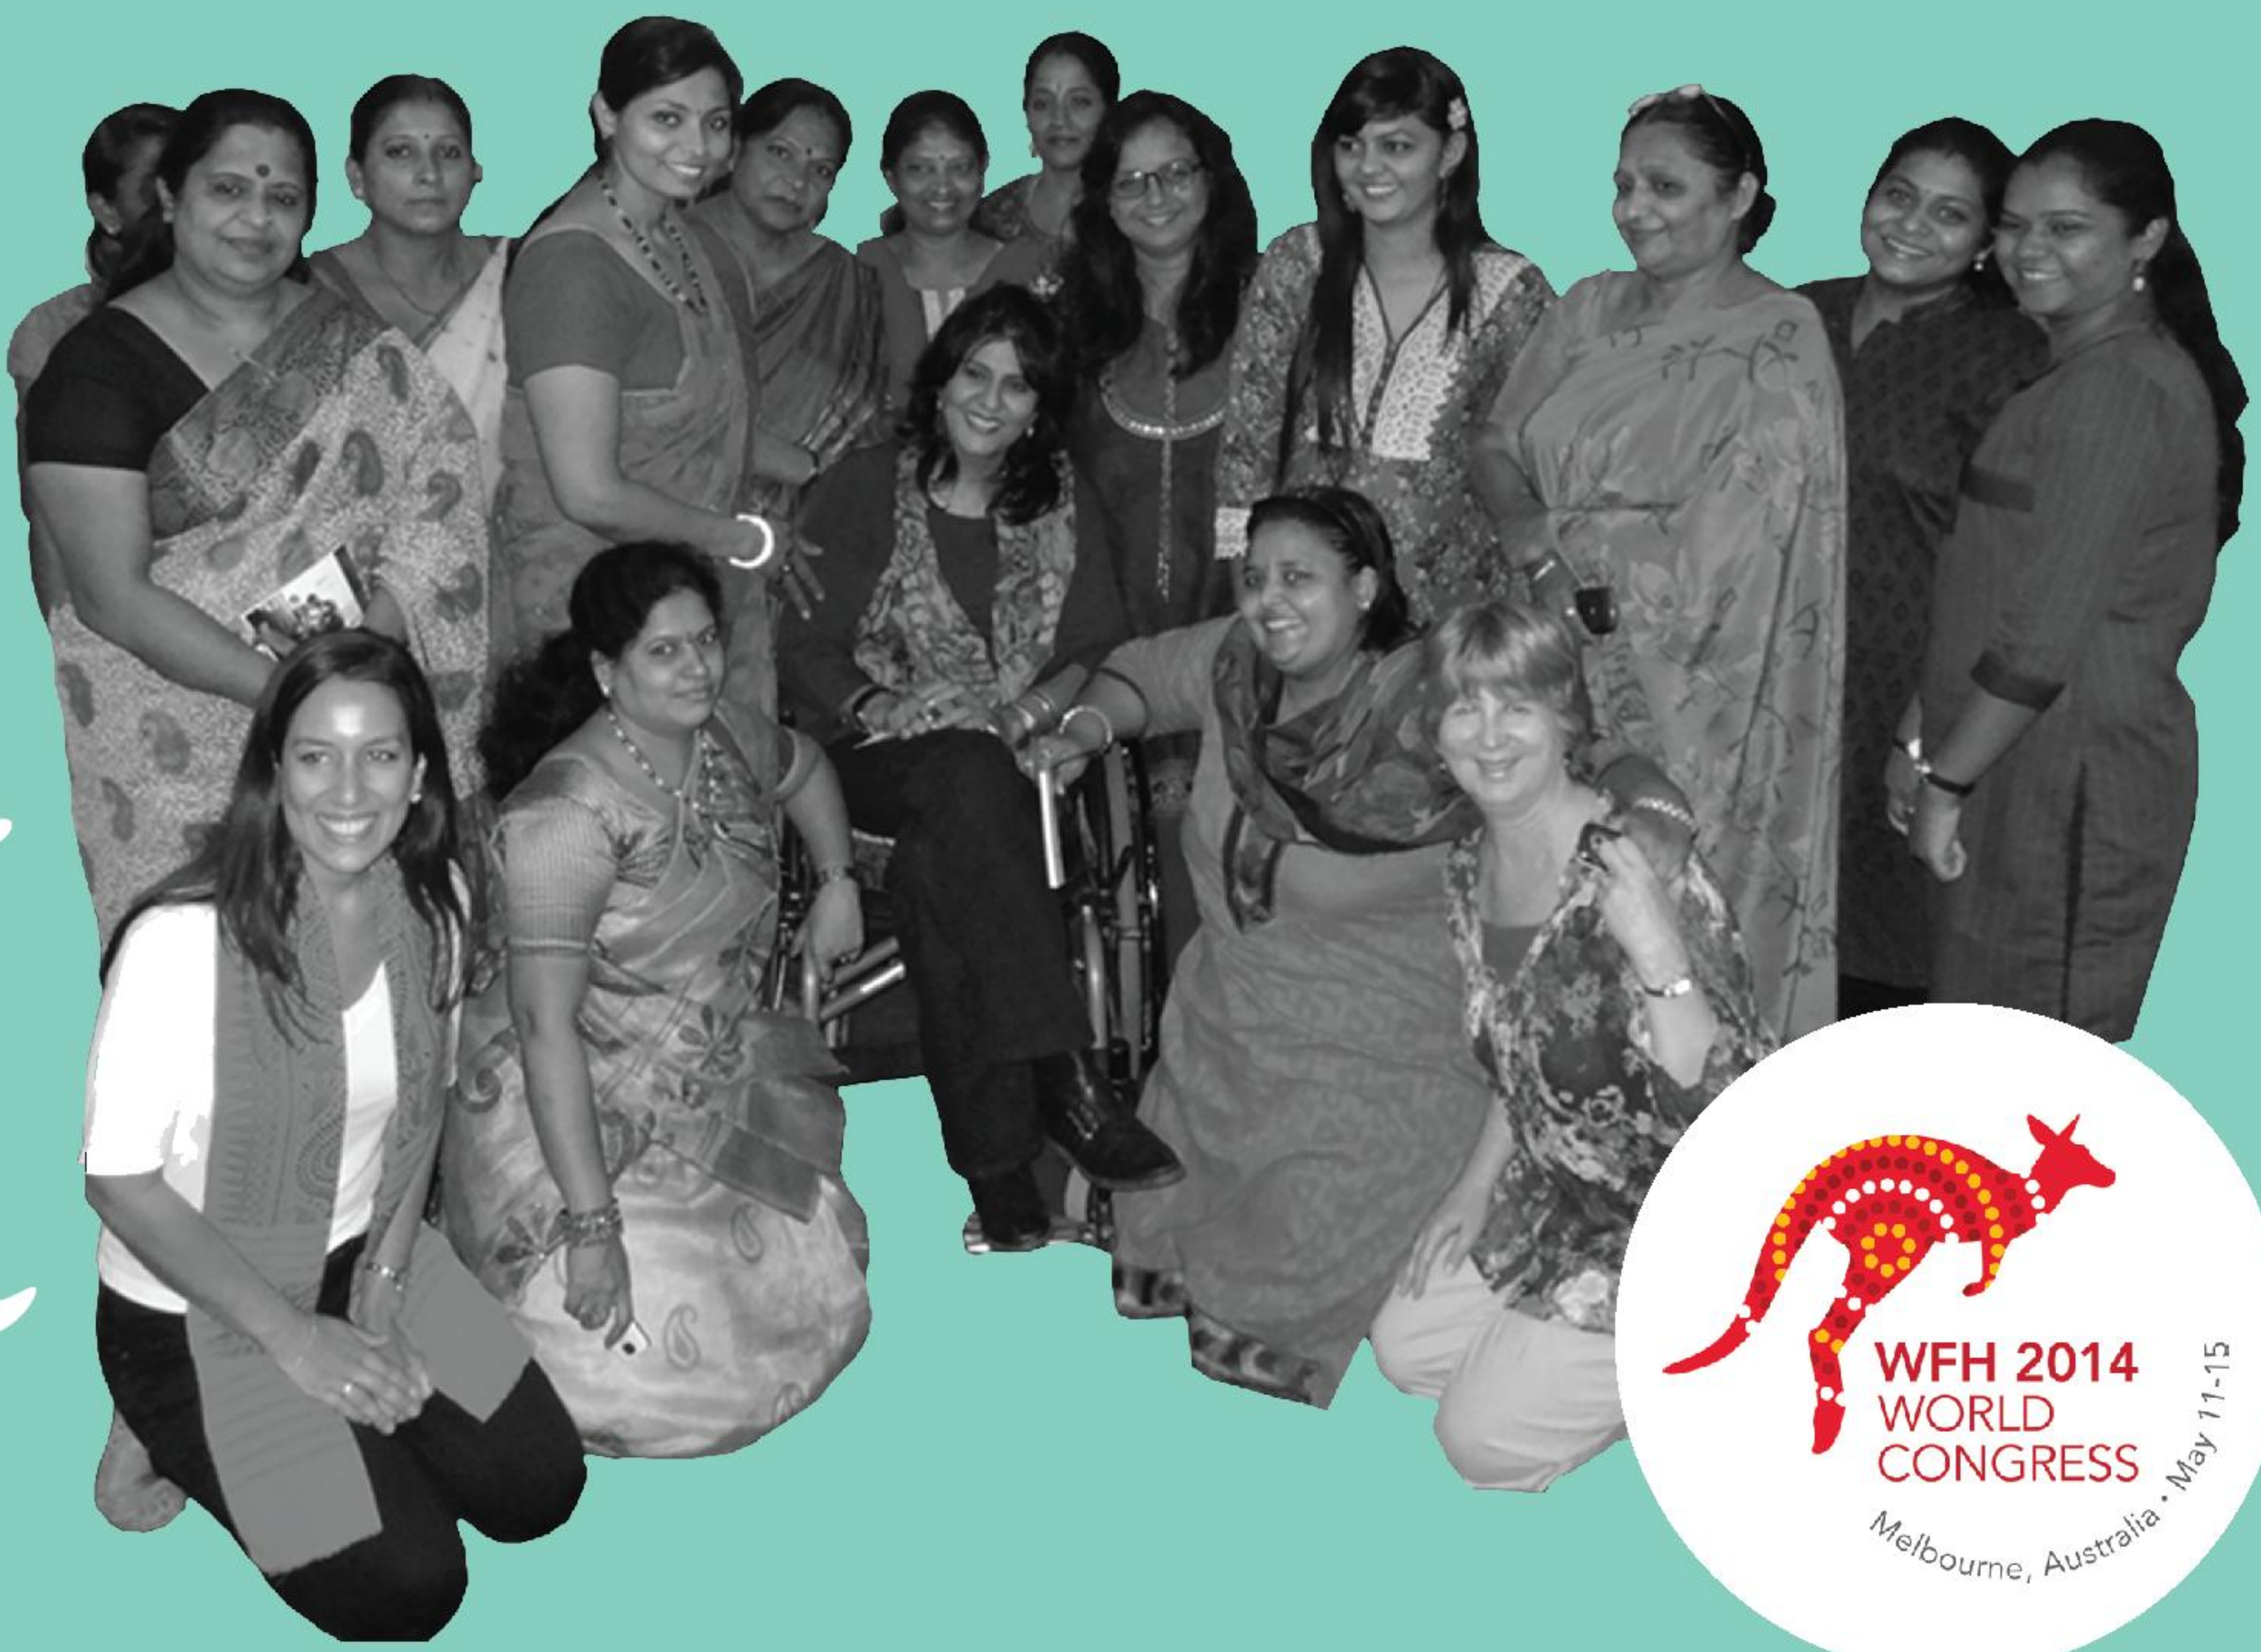

Author - Mukesh Garodia and Indira Nair

Hemophilia Federation (India) with generous funding from Novo Nordisk Haemophilia Foundation

Hemophilia Federation(India)

Hemophilia Without Disablity, Children Free of Pain

novo nordisk haemophilia foundation

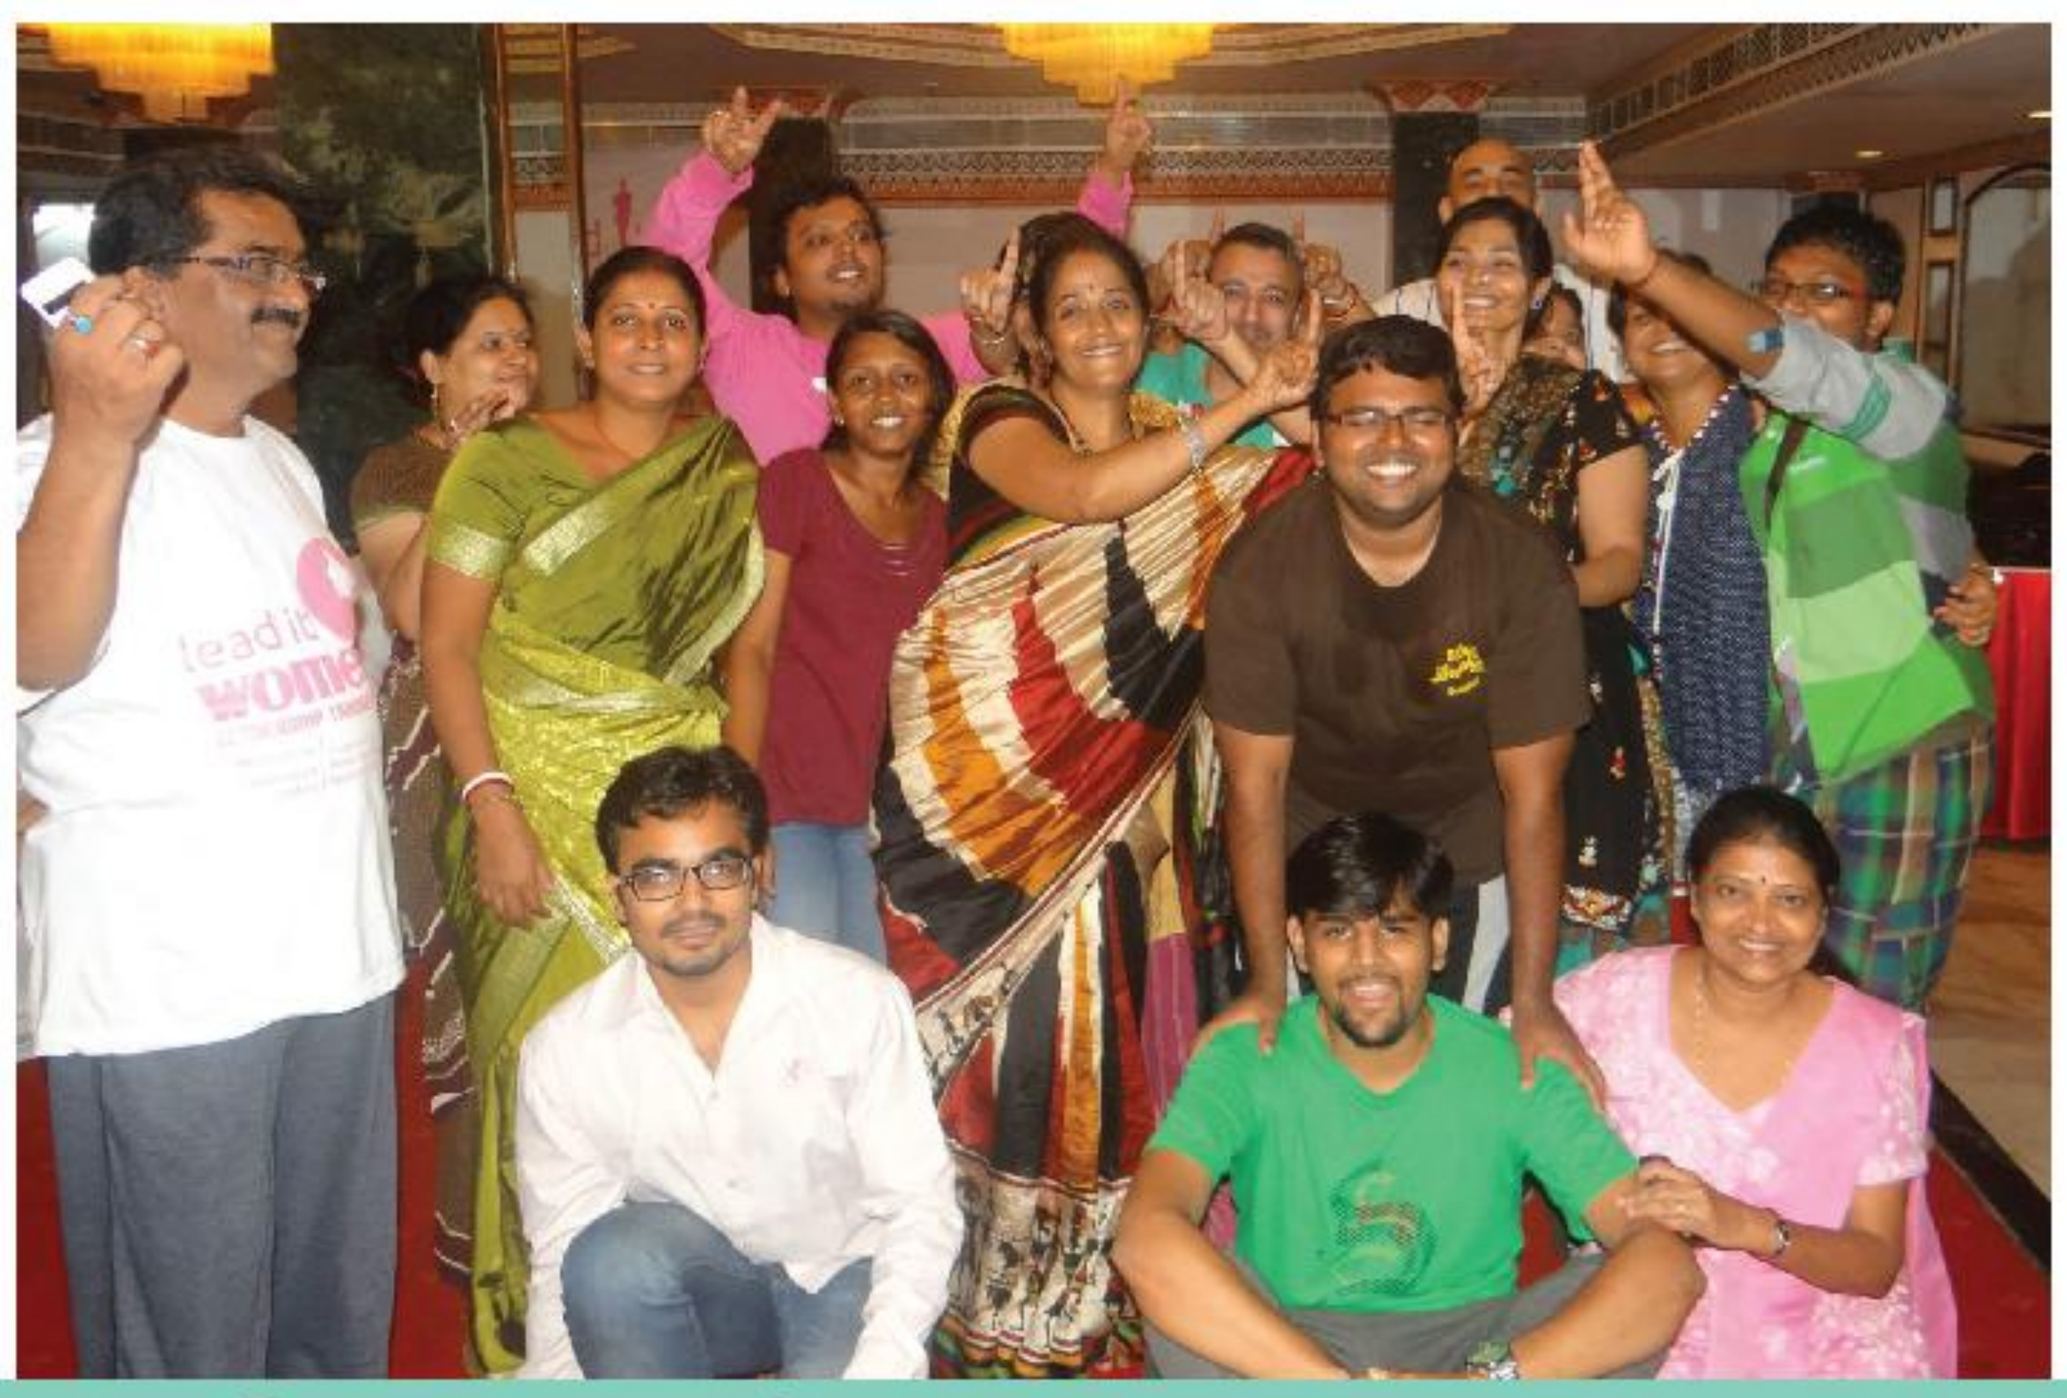

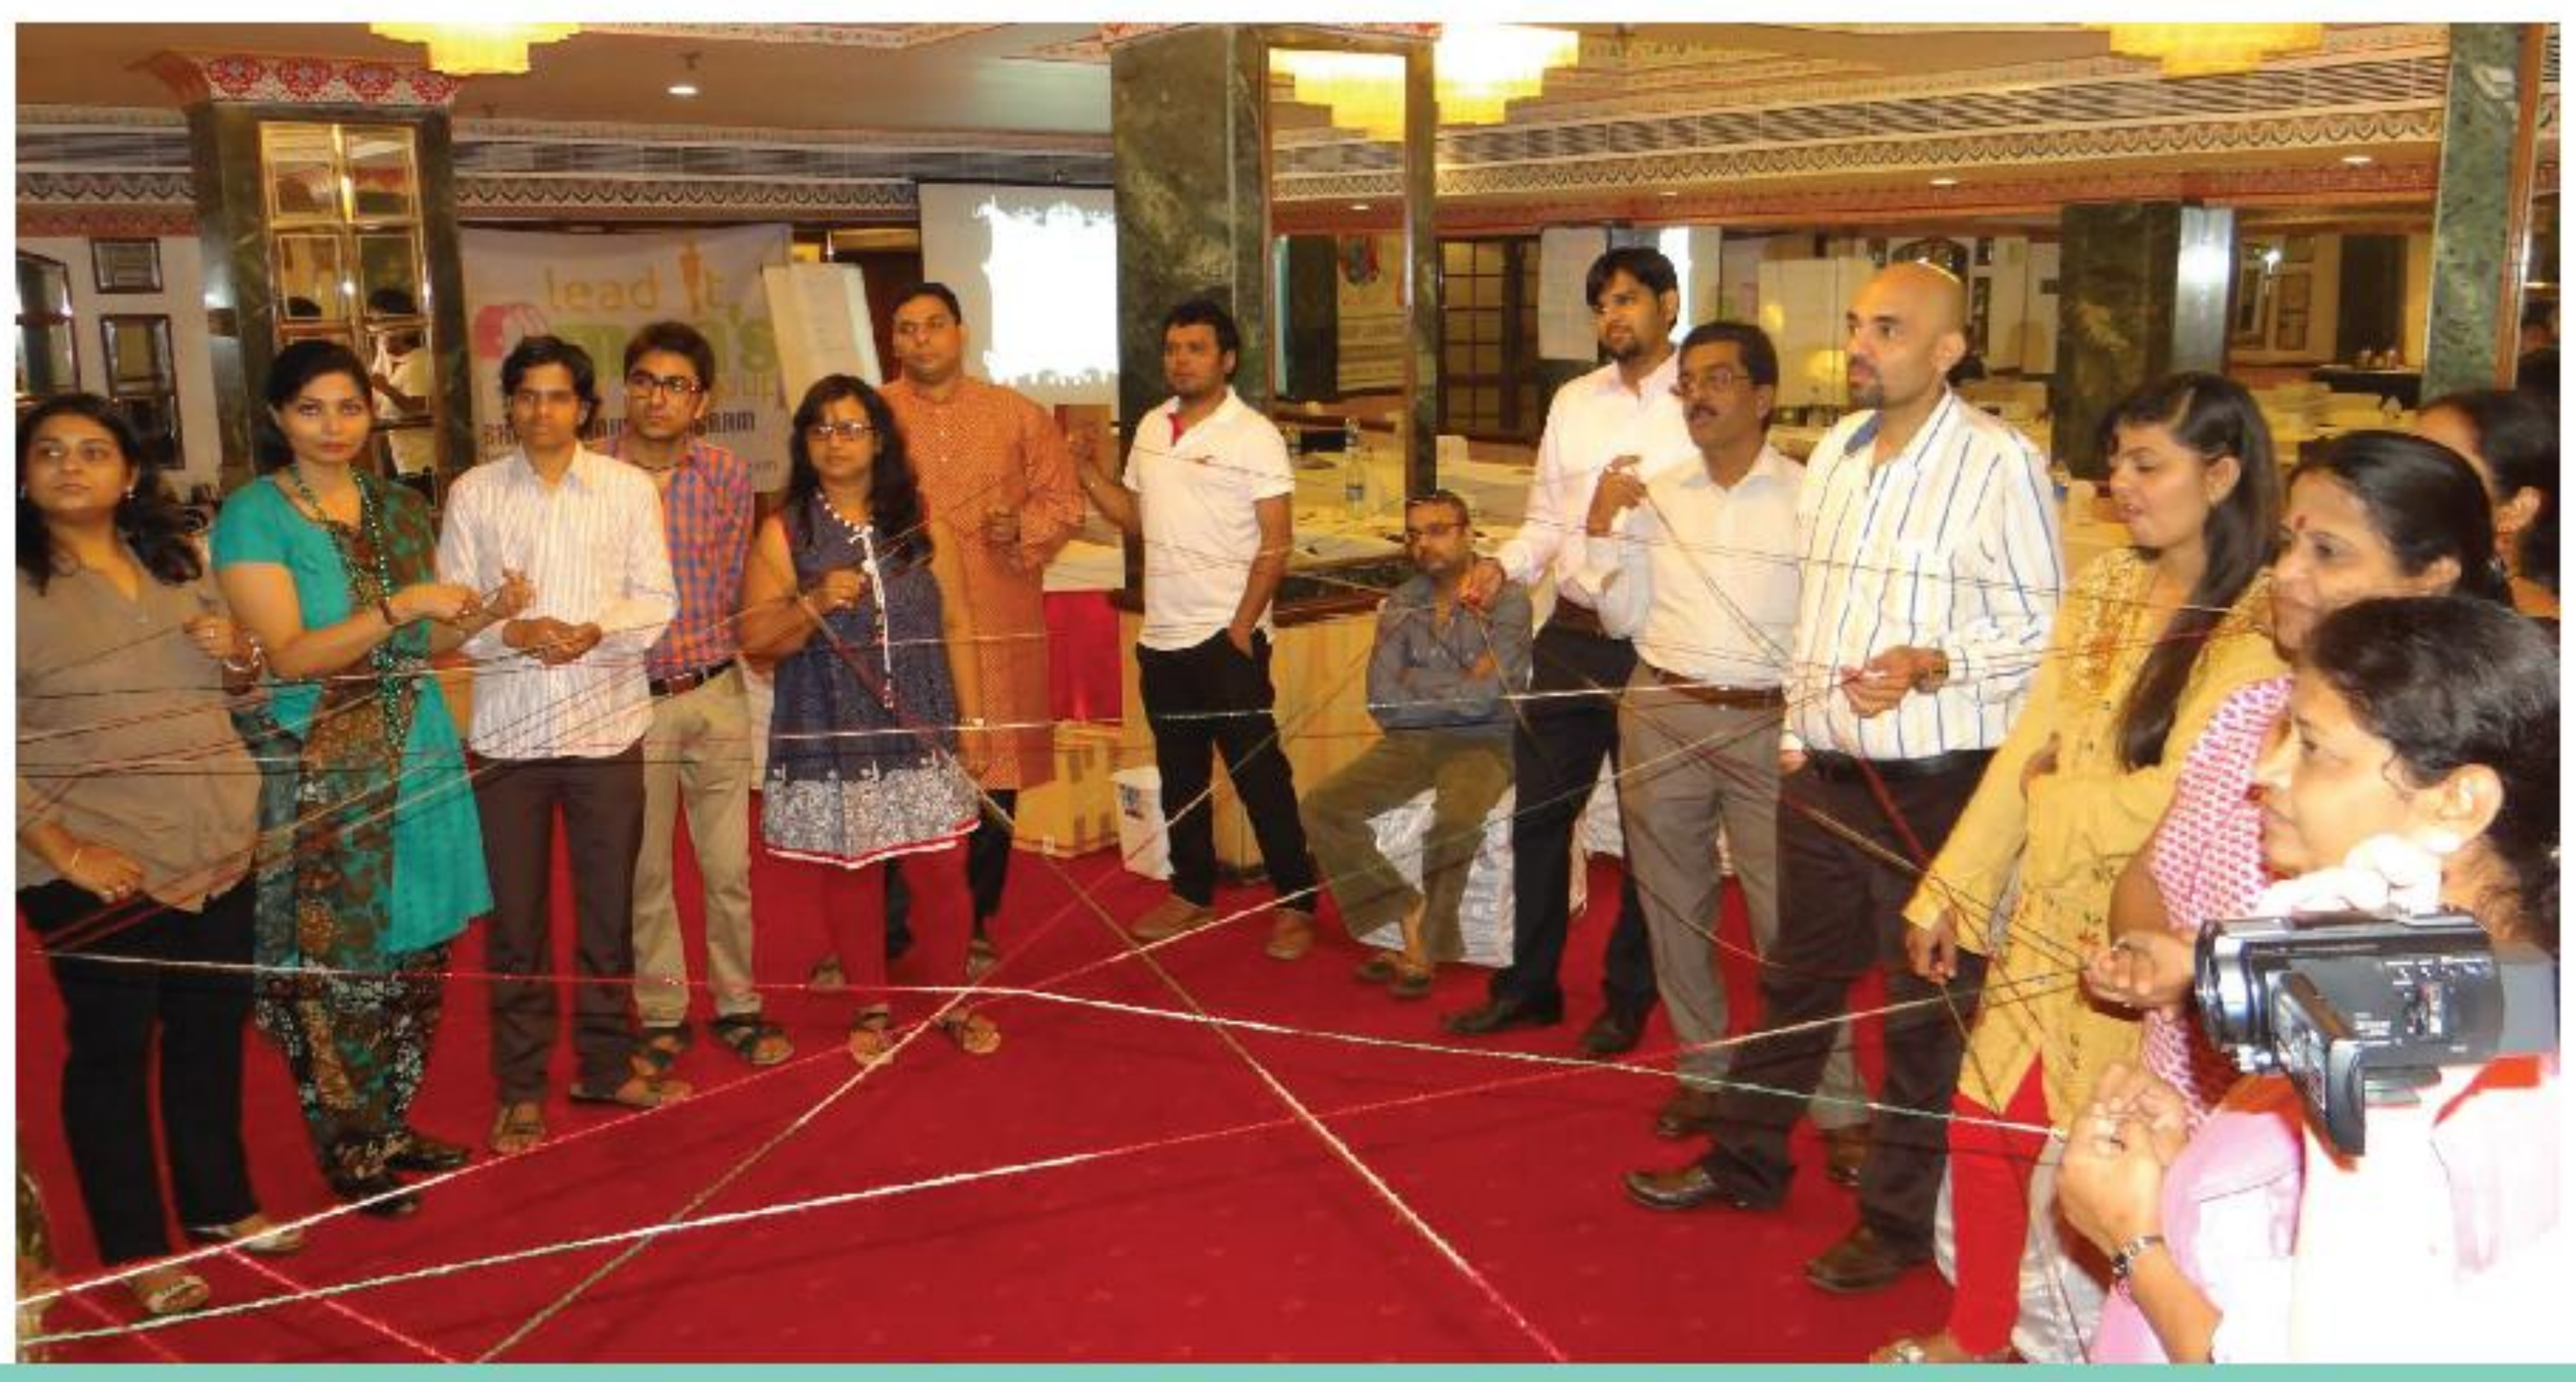

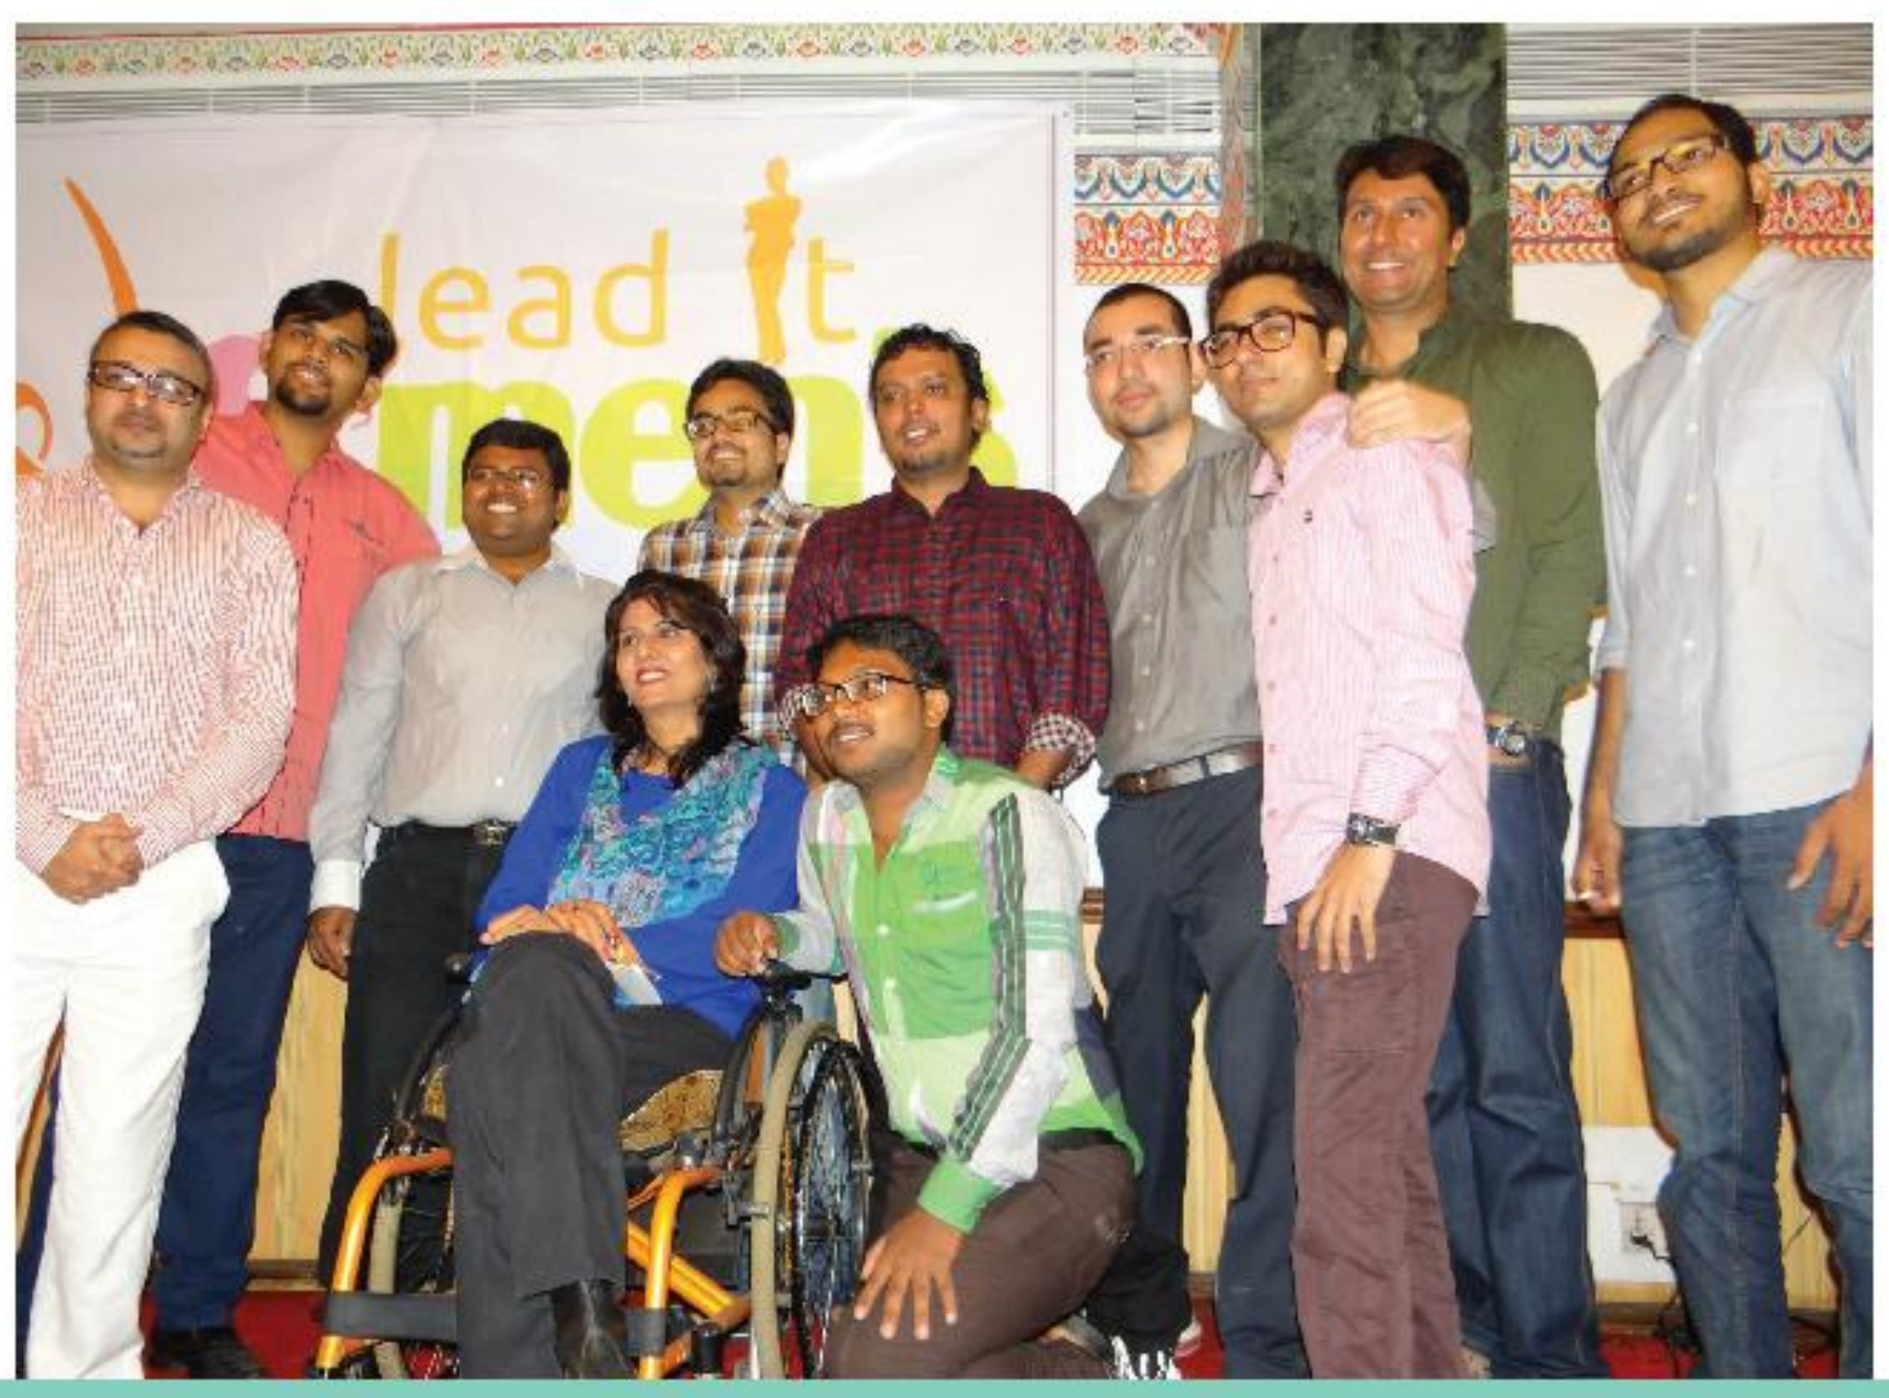

Objectives

Create a platform for a National Women's Group of the Hemophilia Federation of India (HFI)

To provide training on leadership and capacity building, and to provide opportunity for exchange of knowledge, by fostering a global "Sisterhood" amongst the women in the haemophilia community.

Methods

A dozen master trainees will be selected from various chapters in India and trained by experts in key knowledge areas including project management, resource mobilisation, communication, and psycho-social support.

Follow-on efforts will be conducted by these master trainees at chapter and regional levels during workshops and camps, forming new groups and continue development of their existing groups.

A leadership manual for the Women's Group of HFI will be prepared and utilized to ensure continuation of best practices.

Results

A Master Training Programme was launched by the HFI Youth Group Leaders to inaugurate a newly formed Women's Group of HFI.

A chairwoman was elected.

Group mission, values, and objectives were created.

Participants learned basic project management skills to support creation of project plans for their regions.

Issues were raised and discussed.

Conclusions

Women are the wives, mothers, sisters and family members of boys and men who are in the hemophilia community.

A vision for their needs is nonexistent in our worldwide community. They may be carriers (symptomatic or not) and some even have bleeding disorders - or even rare bleeding disorders - yet they must also take care of family members with bleeding disorders, often in isolation.

They would benefit from being a part of a bigger community.

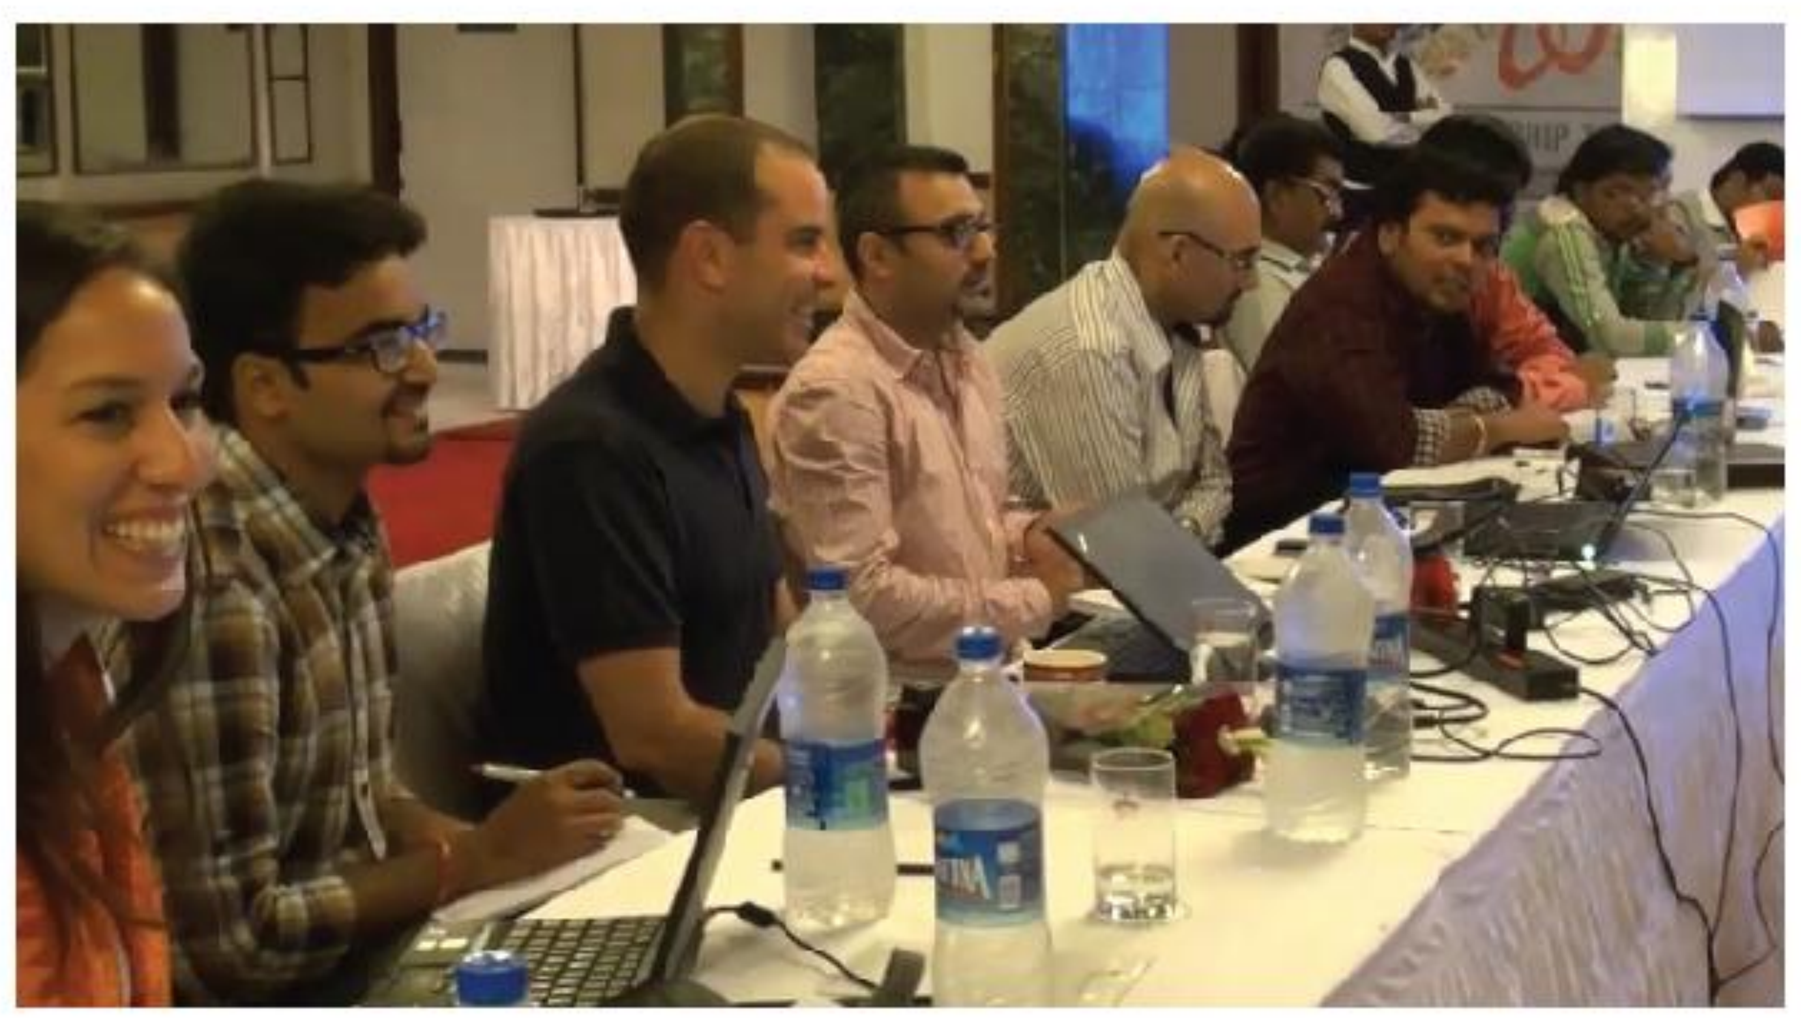

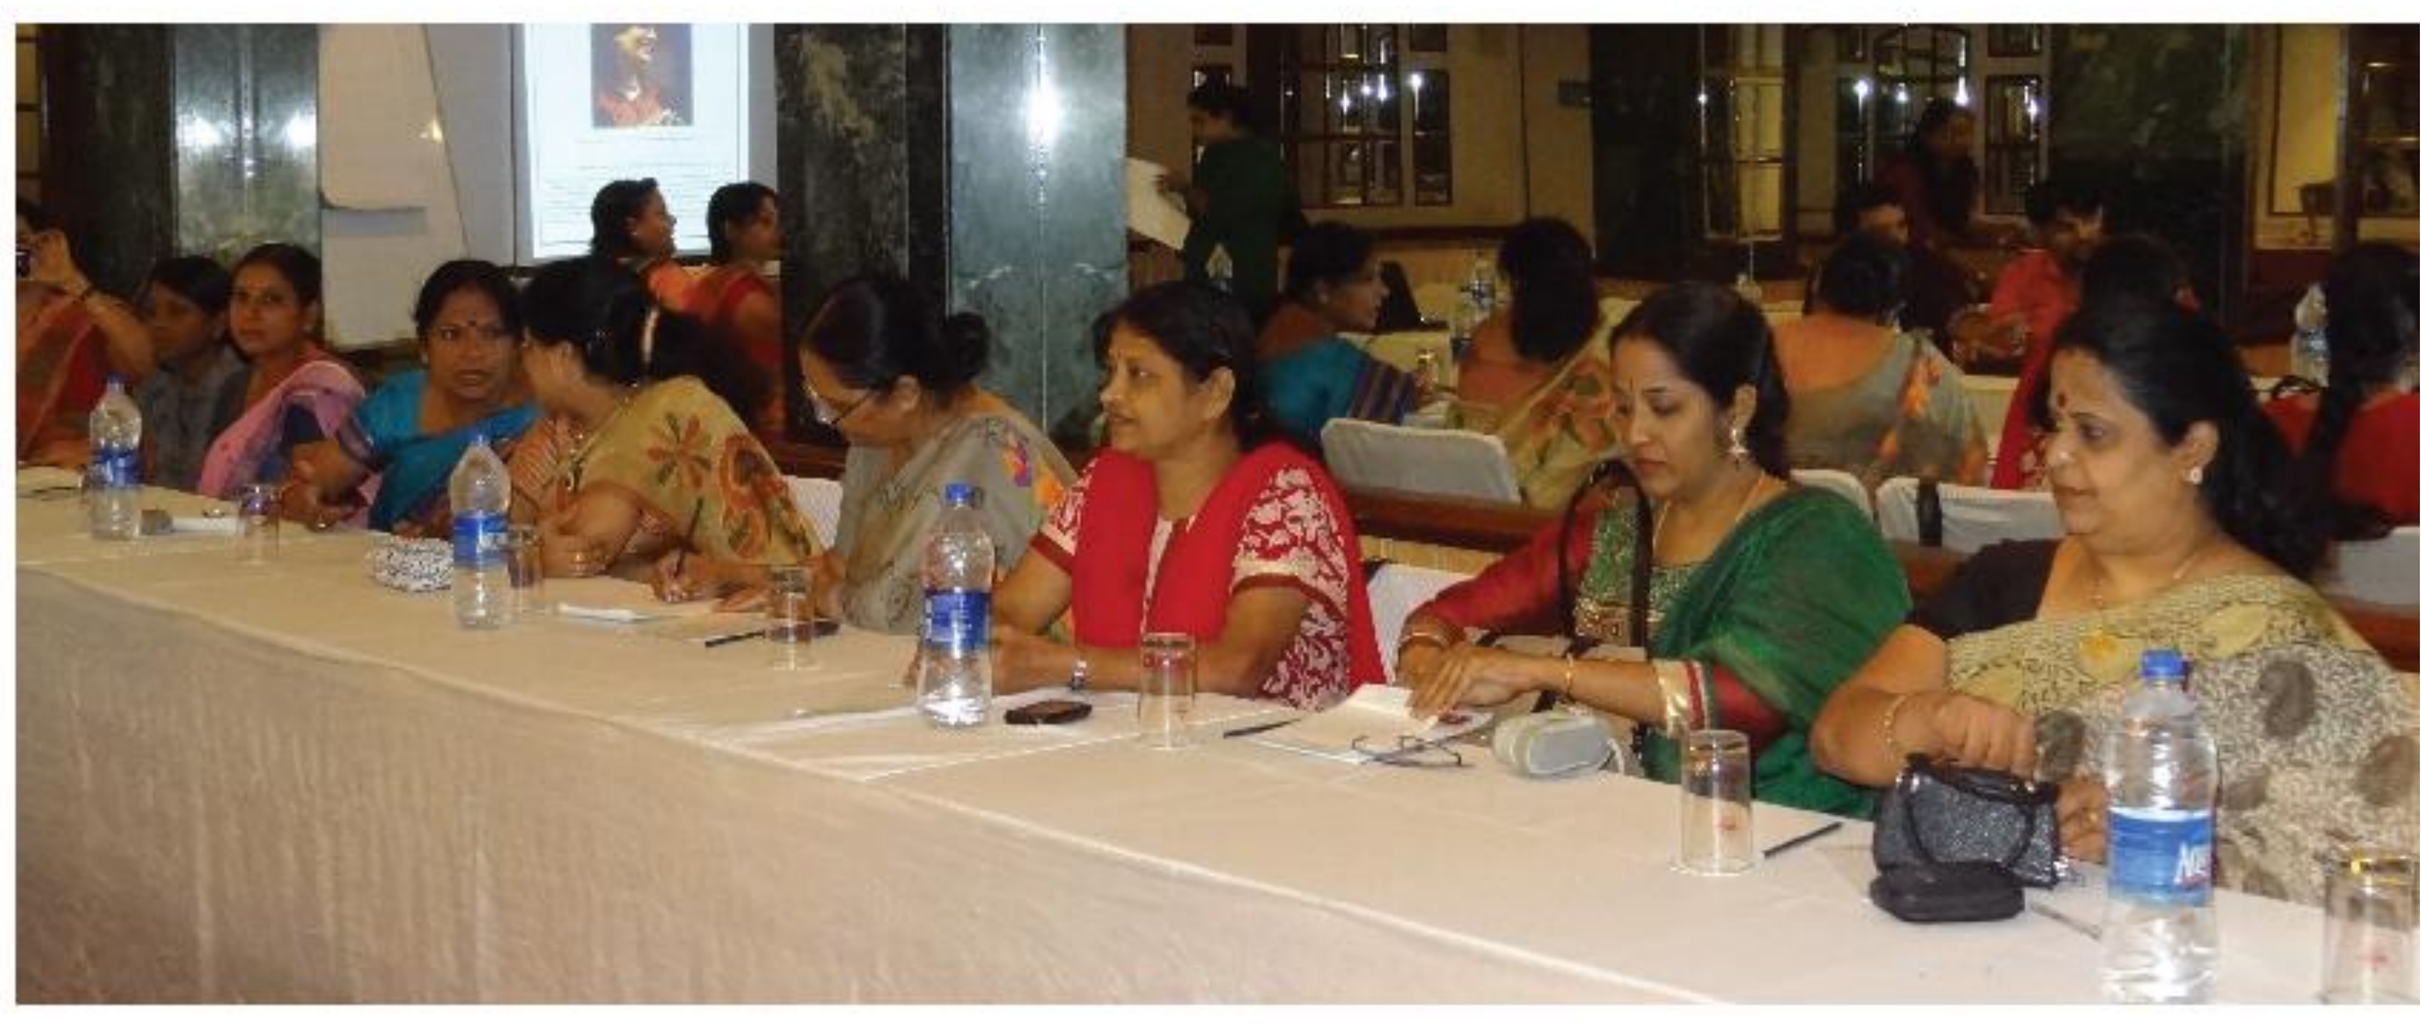

Contribution to the practice of hemophilia and bleeding disorders:

Women leaders and women's groups are in need being developed and sustained but they need a proper platform for operations and sustainability.

A train-the-trainer workshop including empowerment and support from men within their existing hemophilia community promoted the development and sustaining of these women which will strengthen and empower the entire community.

Layout by: Iman Rasheed (imanrshd@gmail.com)

WFH2014

232--P-T

Women and Bleeding Disorders

Mukesh Garodia

DOI: 10.3252/pso.eu.WFH2014.2014

Poster presented at: WFH 2014 World Congress, Melbourne, Australia · May 11-15

PosterSessionOnline.com