Global utilization of the International Prophylaxis Study Group (IPSG) Physical Therapy Hemophilia Joint Health Score (HJHS) Education Web-site Module

Hilliard P[1], Abad A[2], Feldman BM[3], Funk S[5], Manco-Johnson M[5], Petrini P[6], van der Net J[7], Zourikian N[8], and Blanchette VS[4]
on behalf of The International Prophylaxis Study Group

[1]Departments of Rehabilitation Services, Child Health Evaluative Sciences[2], Pediatrics, Division of Rheumatology[3] and Hematology/Oncology[4], The Hospital for Sick Children, University of Toronto, Toronto, ON, Canada; [5] University of Colorado, Hemophilia & Thrombosis Center, Denver, Colorado, USA; [6]Departments of Pediatrics, Division of Blood Coagulation Disorders, Karolinska University Hospital, Stockholm, Sweden; [7]Child Development Exercise Center, Division of Pediatrics, University Children's Hospital and Medical Center at Utrecht, The Netherlands; [8]Department of Physiotherapy, Sainte-Justine University Hospital Center, Montreal, QC, Canada

INTRODUCTION & OBJECTIVES

- The International Prophylaxis Study Group (IPSG) is a not-for-profit collaborative group of health care professionals involved with the assessment and care of individuals with inherited bleeding disorders (focus on hemophilia), and with a special interest in long-term regular replacement therapy (prophylaxis).
- An IPSG website was launched in February 2013 to increase global awareness about the activities of the International Prophylaxis Study Group and disseminate knowledge and information about outcome measurement tools developed by the Expert Working Groups.
- The IPSG Physical Therapy Expert Working Group, formed in 2002, developed and tested the Hemophilia Joint Health Score (HJHS), a scoring tool for the evaluation of musculoskeletal impairment. The HJHS was initially developed to help capture early clinical signs of joint disease in children and is currently being studied in other populations.

EDUCATION MATERIAL

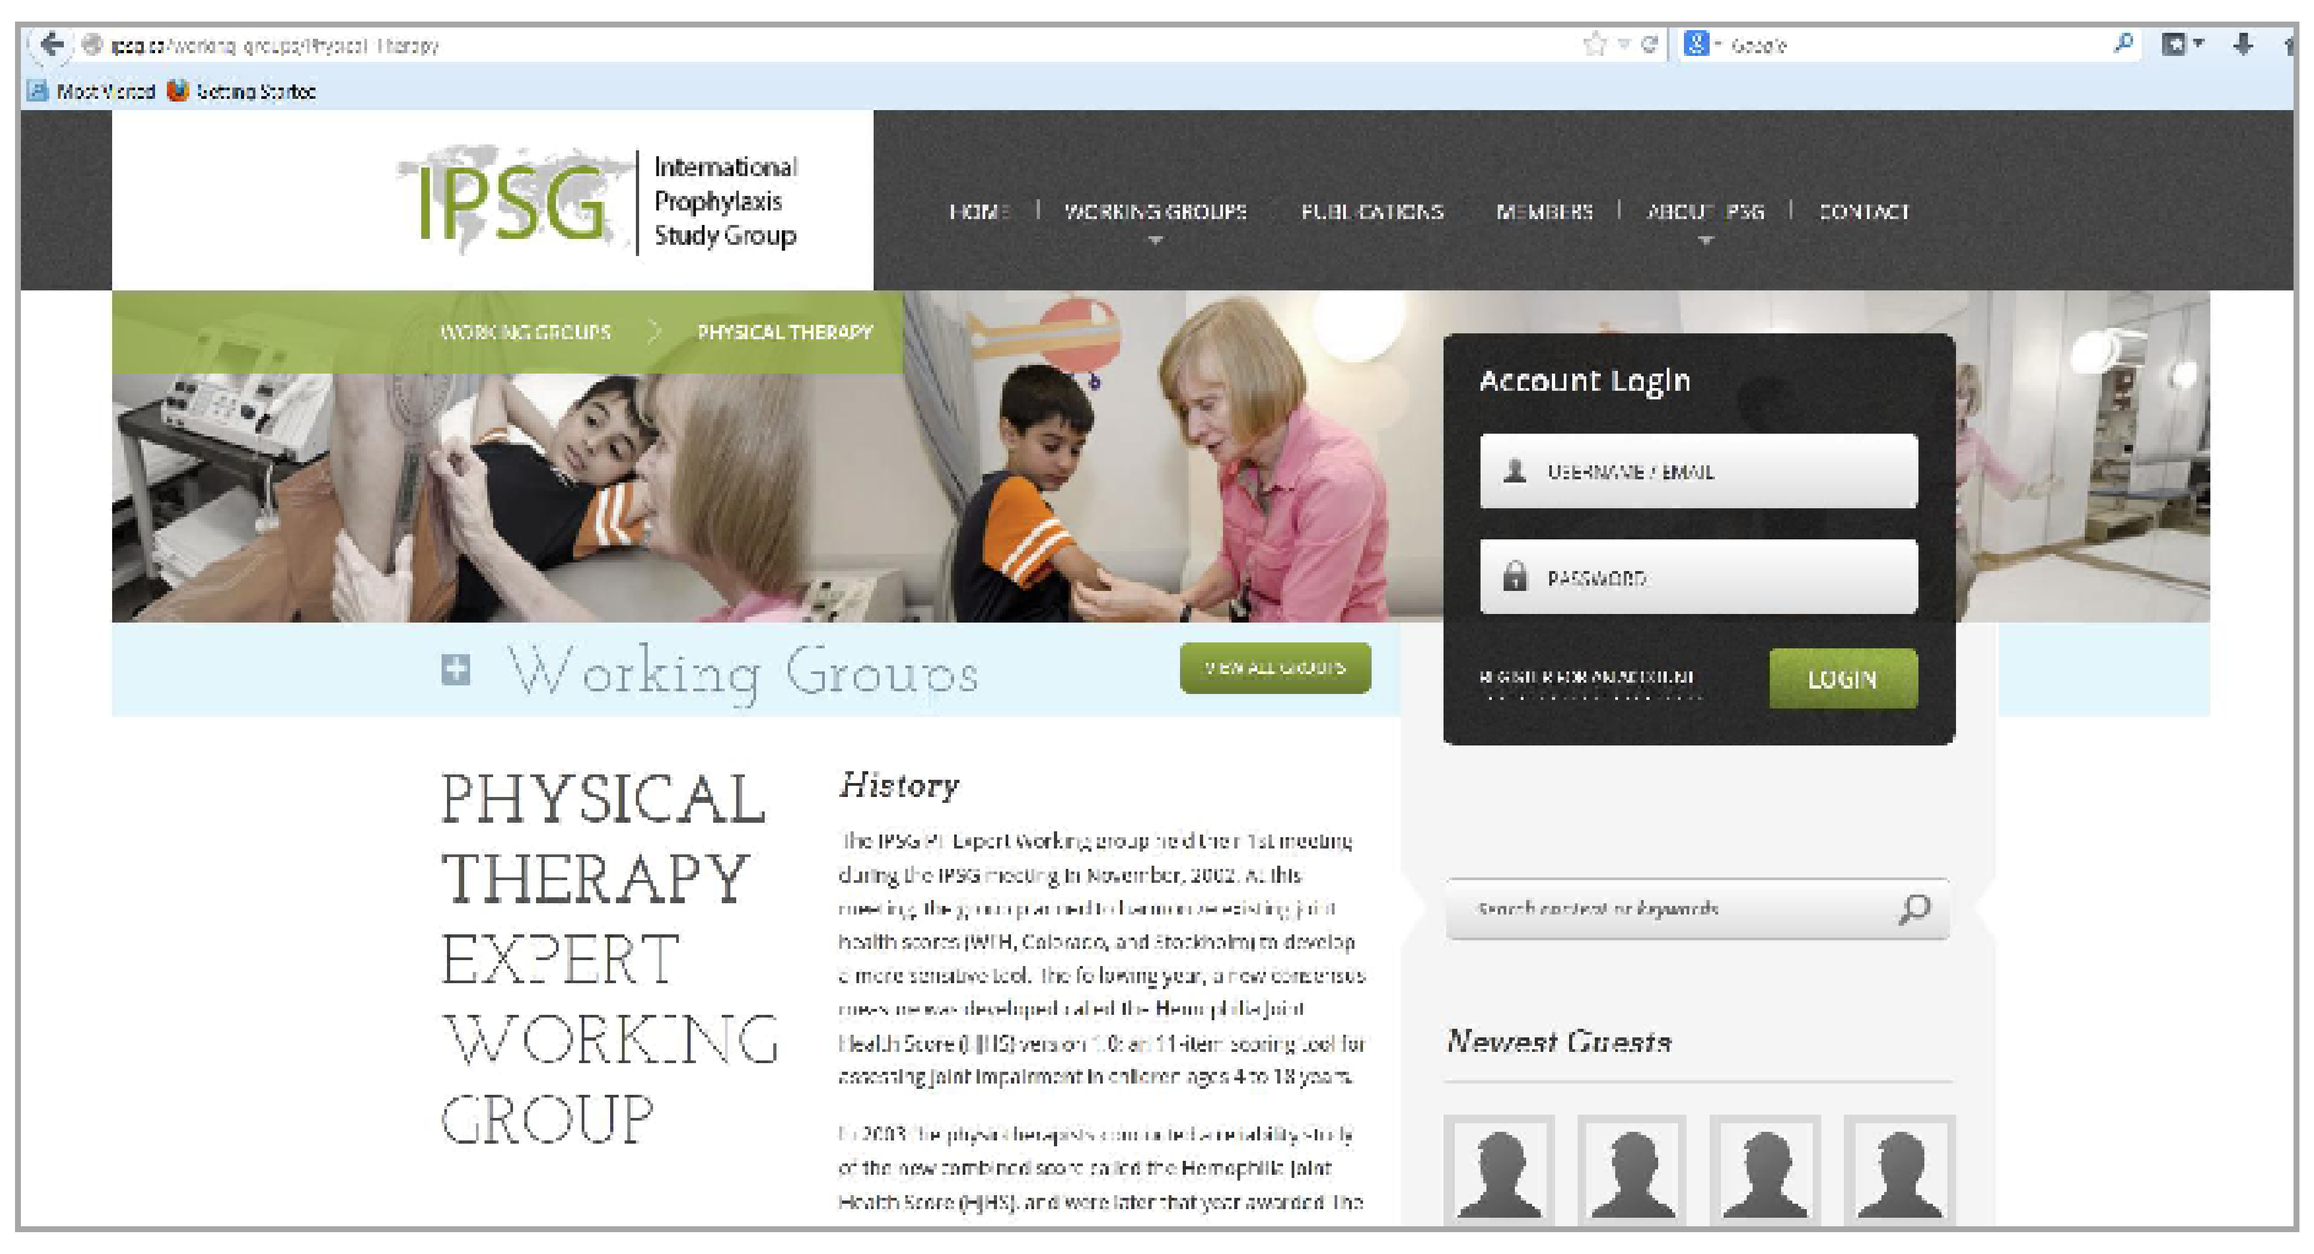

IPSG International Prophylaxis Study Group
HOME | WORKING GROUPS | PUBLICATIONS | MEMBERS | ABOUT IPSG | CONTACT
Account Login
LOGIN
Working Groups
PHYSICAL THERAPY EXPERT WORKING GROUP
History
Newest Guests

- The Physical Therapy module, the initial Expert Working Group component of the web-site, is comprised of the original English HJHS score sheet, worksheets and instruction manual and has been translated into French, German, Spanish and Simplified Chinese.
- An English language HJHS instructional video can be downloaded or streamed for viewing and has been translated into French, German, and Simplified Chinese (Mandarin) using systematic processes to ensure technical accuracy and content relevance. Spanish and Portuguese translations are currently in progress.

Country	Language	HJHS 2.1 Instruction Manual (Includes worksheets & Score sheet)	HJHS 2.1 Instructional Video
Canada	English	✓	✓
France	French	✓	✓
Germany	German	✓	✓
Spain	Spanish	✓	In progress
China	Simplified Chinese (Mandarin)	✓	✓
Brazil	Portuguese	In progress	In progress

DEMOGRAPHICS & REQUESTS

- Visitors from a total of 84* countries over 6 continents have visited the web-site since February 2013
- Unique web-site visits (unduplicated) = 1,520
- Total number of registrants for the IPSG web-site = 270

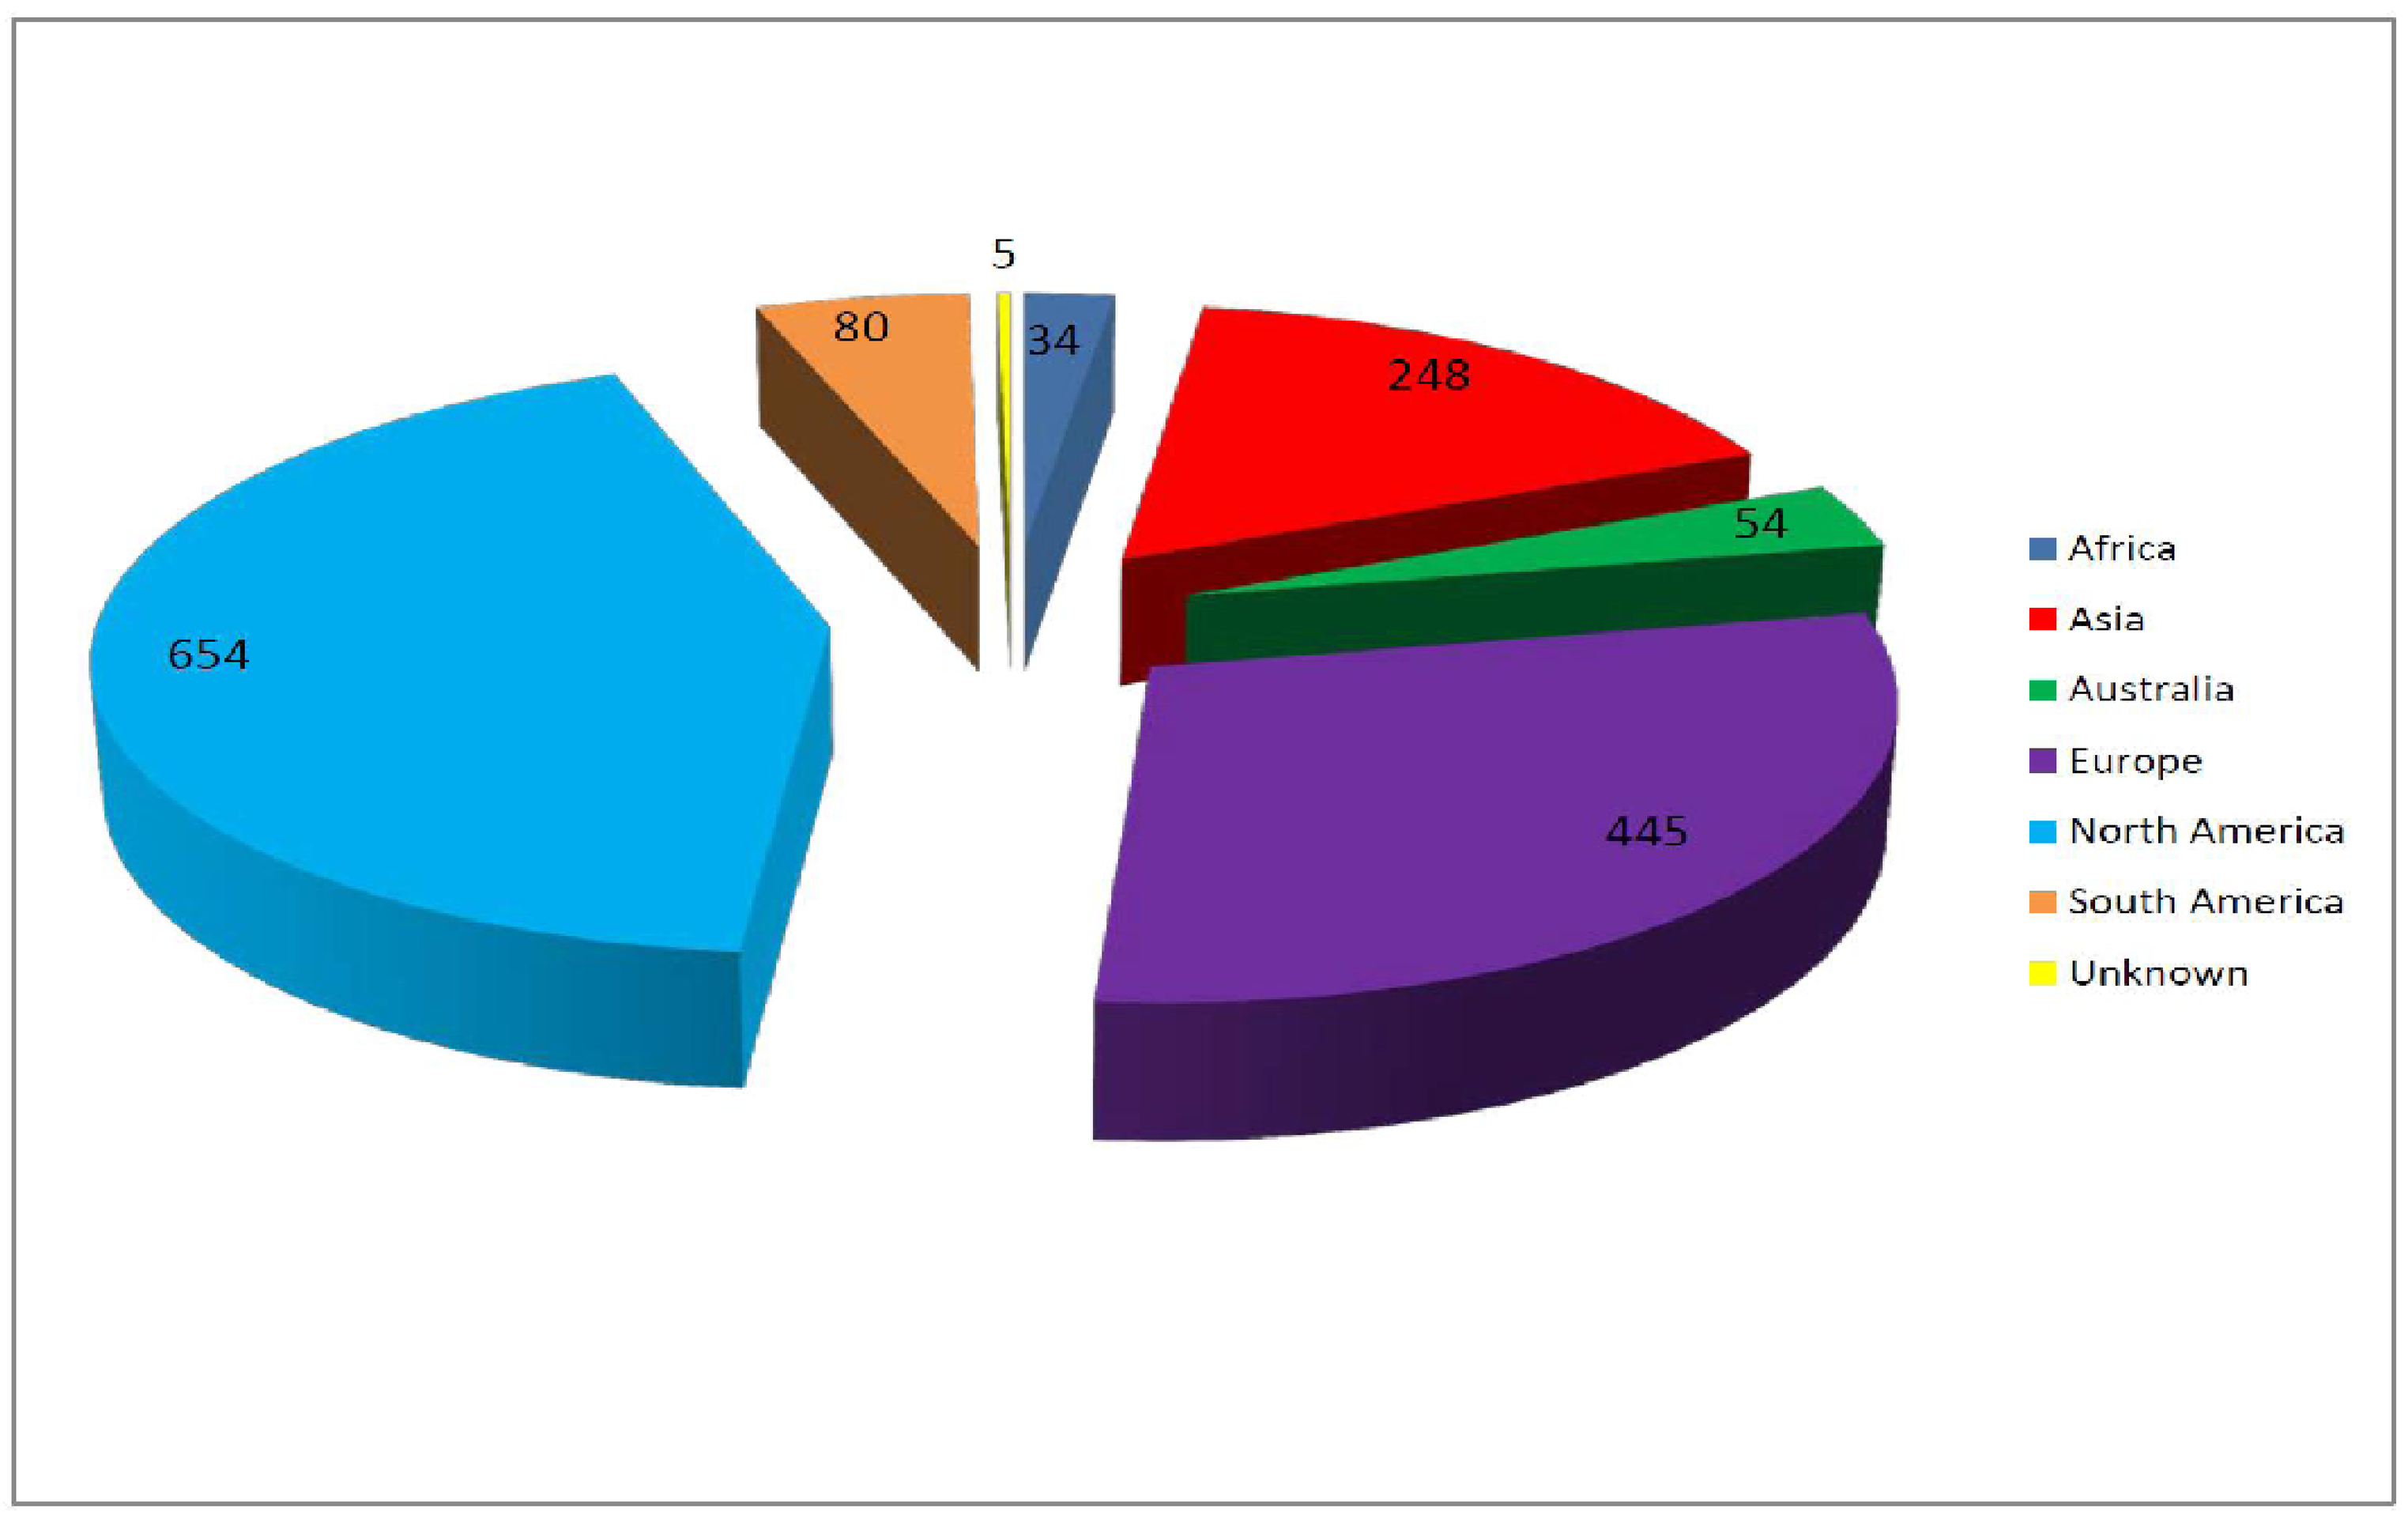

5
80
34
248
54
654
445
Africa
Asia
Australia
Europe
North America
South America
Unknown

* Algeria, Argentina, Australia, Austria, Bahrain, Barbados, Belgium, Bolivia, Bosnia and Herzegovina, Brazil, Bulgaria, Cameroon, Canada, Chile, China, Colombia, Costa Rica, Czech Republic, Denmark, Egypt, Estonia, Estonia, Finland, France, Germany, Ghana, Greece, Guam, Hong Kong, Hungary, India, Indonesia, Iran, Ireland, Israel, Italy, Japan, Kenya, Kyrgyzstan, Latvia, Lebanon, Lithuania, Luxembourg, Malaysia, Mauritius, Mexico, Moldova, Morocco, Netherlands, New Zealand, Nigeria, Norway, Oman, Pakistan, Peru, Philippines, Poland, Portugal, Qatar, Romania, Russia, Rwanda, Saudi Arabia, Serbia, Singapore, Slovakia, Slovenia, South Africa, South Korea, Spain, Sri Lanka, Suriname, Sweden, Switzerland ,Taiwan, Tunisia ,Turkey ,United Arab Emirates ,United Kingdom ,United States , Venezuela, Vietnam

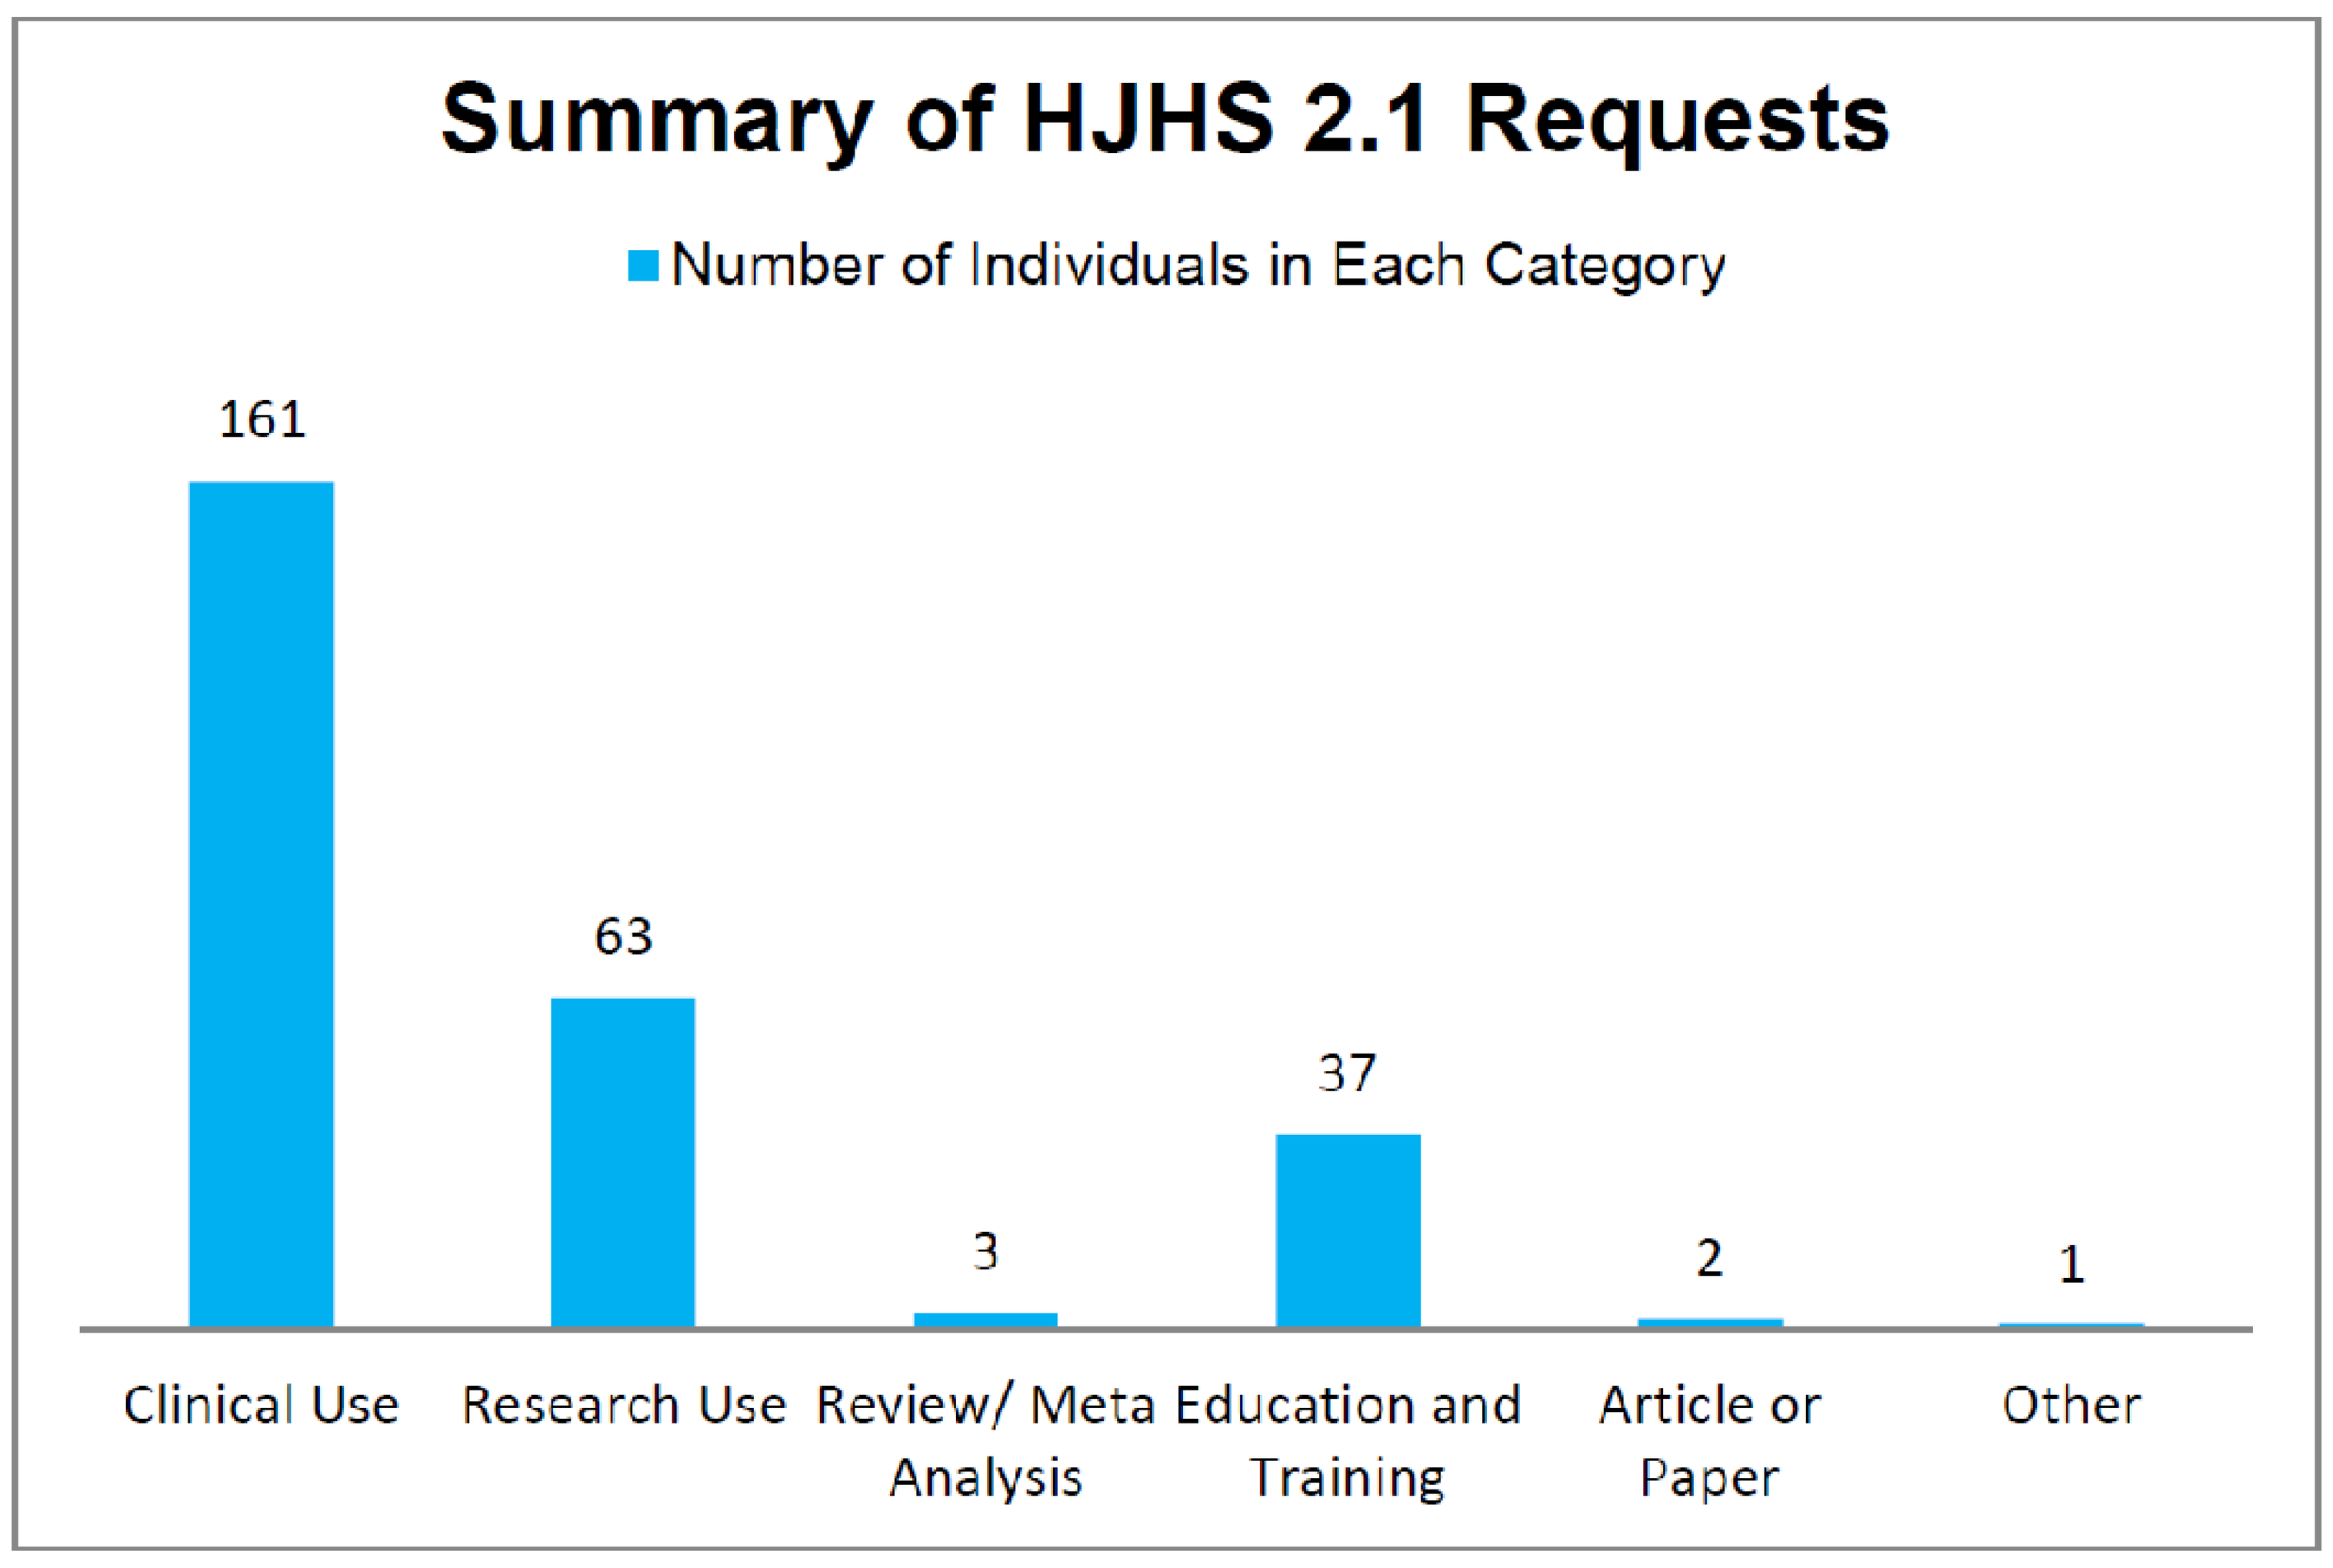

Summary of HJHS 2.1 Requests
Number of Individuals in Each Category
161
63
3
37
2
1
Clinical Use
Research Use
Review/ Meta Analysis
Education and Training
Article or Paper
Other

SUMMARY

- **The IPSG website continues to contribute to the education of health professionals involved in the care of patients with hemophilia and other bleeding disorders worldwide.**
- **For more information please visit: www.ipsg.ca**

ACKNOWLEDGEMENTS

We would like to thank the patients/families who participated in the production of the video, the healthcare experts who reviewed the education material, Associated Producers Ltd. and Therefore Interactive.

We would like to express our thanks to the International Prophylaxis Study Group.

The development of the web-site was supported by unrestricted grants to the International Prophylaxis Study Group (IPSG) from Bayer Healthcare; Baxter BioScience; CSL Behring; Pfizer; and Novo Nordisk Healthcare AG; and was administered through The Hospital for Sick Children Foundation, Toronto, Canada

WFH2014 143--P-T
Educational Models
Pamela Hilliard
Poster presented at: