Max the frog: an educative tool for toddlers with hemophilia

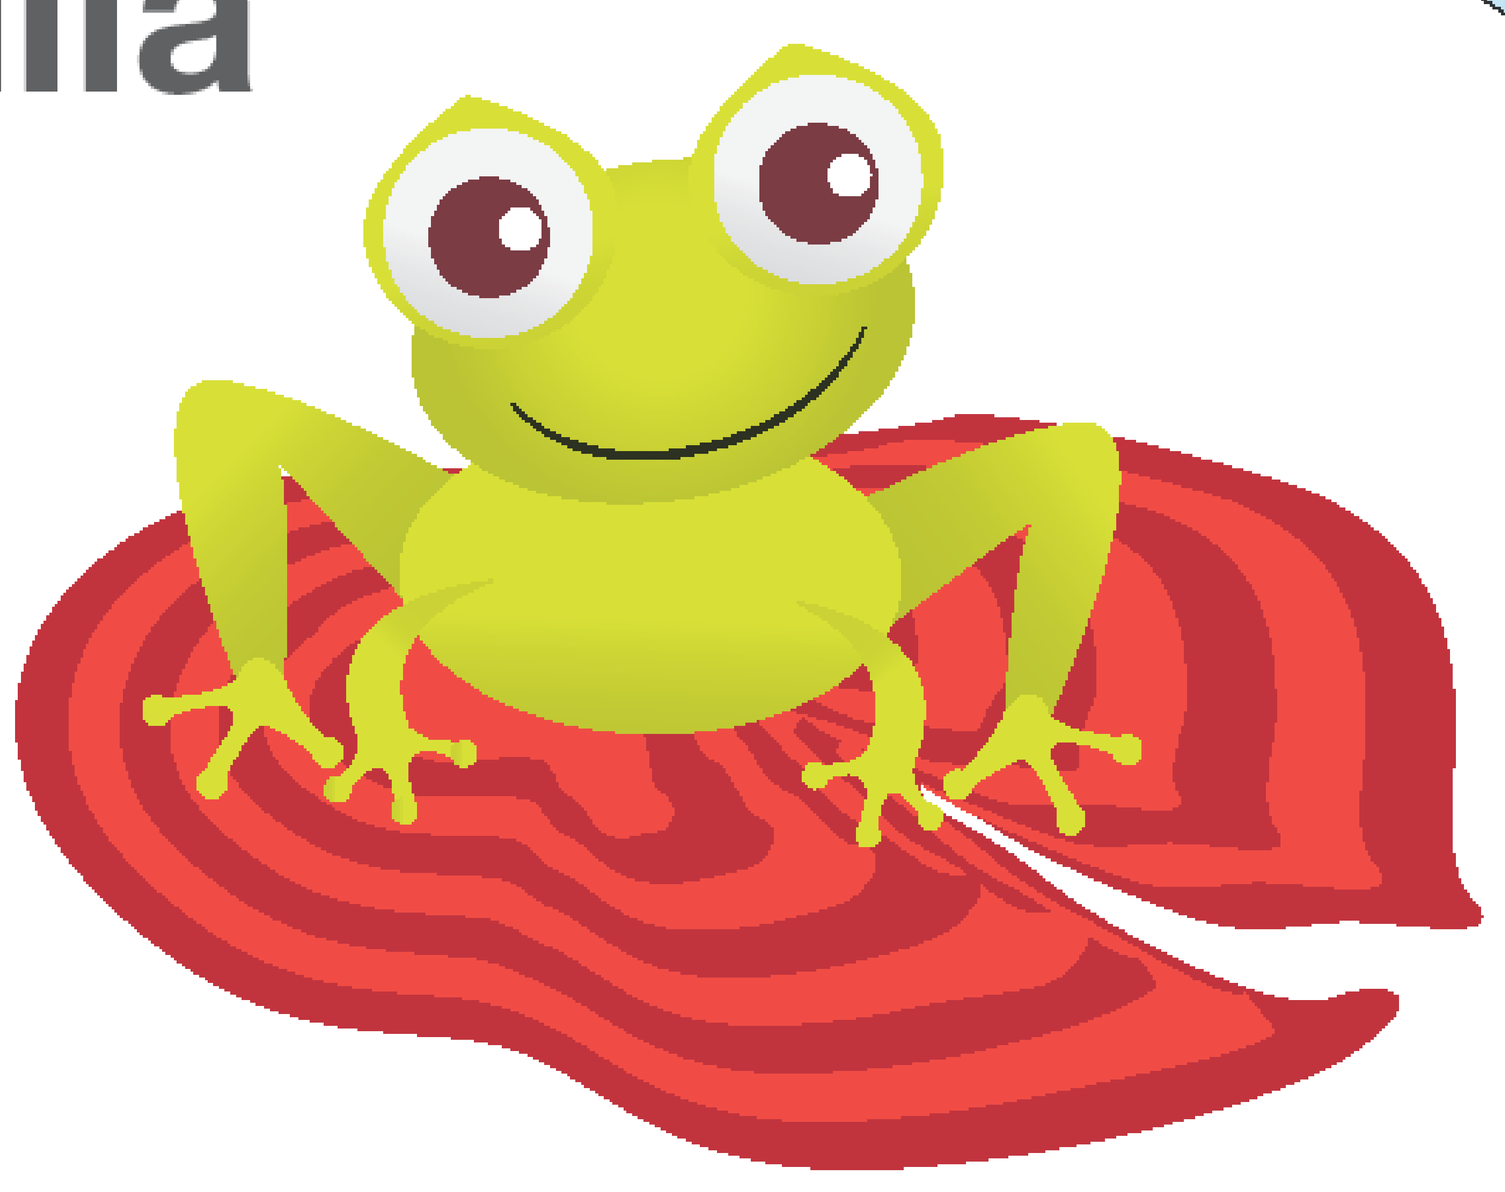

F. Delacollette[1], L. Leenders [2], C. Moris [3]

[1] **Pediatric Home Care, CHR Citadelle Liège, Belgium**

[2] **Hemato-Oncology Unit, University Children Hospital Queen Fabiola (HUDERF), Brussels, Belgium**

[3] **Bayer Belgium**

Background

Severe hemophilia is characterized by recurrent, and often spontaneous, bleedings into muscles and joints from an early age of life. The consequences of such bleedings are progressive joint damage, leading to arthropathy associated with long-term morbidity and severe physical and psychosocial impairment.

The challenge is to optimize the effective strategy to prevent joint bleeding, by improving our knowledge of the disease. The therapeutic education of patients (TEP) permits a sick person to acquire the essential skills needed to live with their specific chronic illness. The patient is considered autonomous, not just capable of making his or her own health-related decisions, but a full partner in the process of providing care.

TEP confirms the practical utility of knowledge and skills acquired and implemented in the patients' daily lives.

Objectives

Children are exposed very early to frequent injections but do they know the goal / benefit?

This tool aims to teach young children with hemophilia about one of the most important aspects of their illness, namely the importance of regular treatment and compliance.

The illustrated story of "**Max the frog**" allows the child to connect with the reality of his illness. This tool enables us, after reading, to begin a structured discussion with the child in order to make the connection between storytelling and his real life. The questions asked after reading will give to the child the opportunity to freely express his thoughts and perceptions.

The goal is to translate the benefits of regular treatment in a fantasy world. The child understands that his treatment is really positive and that he has to practice it in his daily life. The unpleasant injection allows him to do things he could perhaps not do otherwise. If he becomes aware of the importance of prophylactic treatment, it will be easier to integrate this into his daily life.

The story does not aim to assess the knowledge of the child about his illness in general at the beginning. This assessment can be done during the discussion following the reading of the story. In this case, the questions guiding the conversation will be oriented differently.

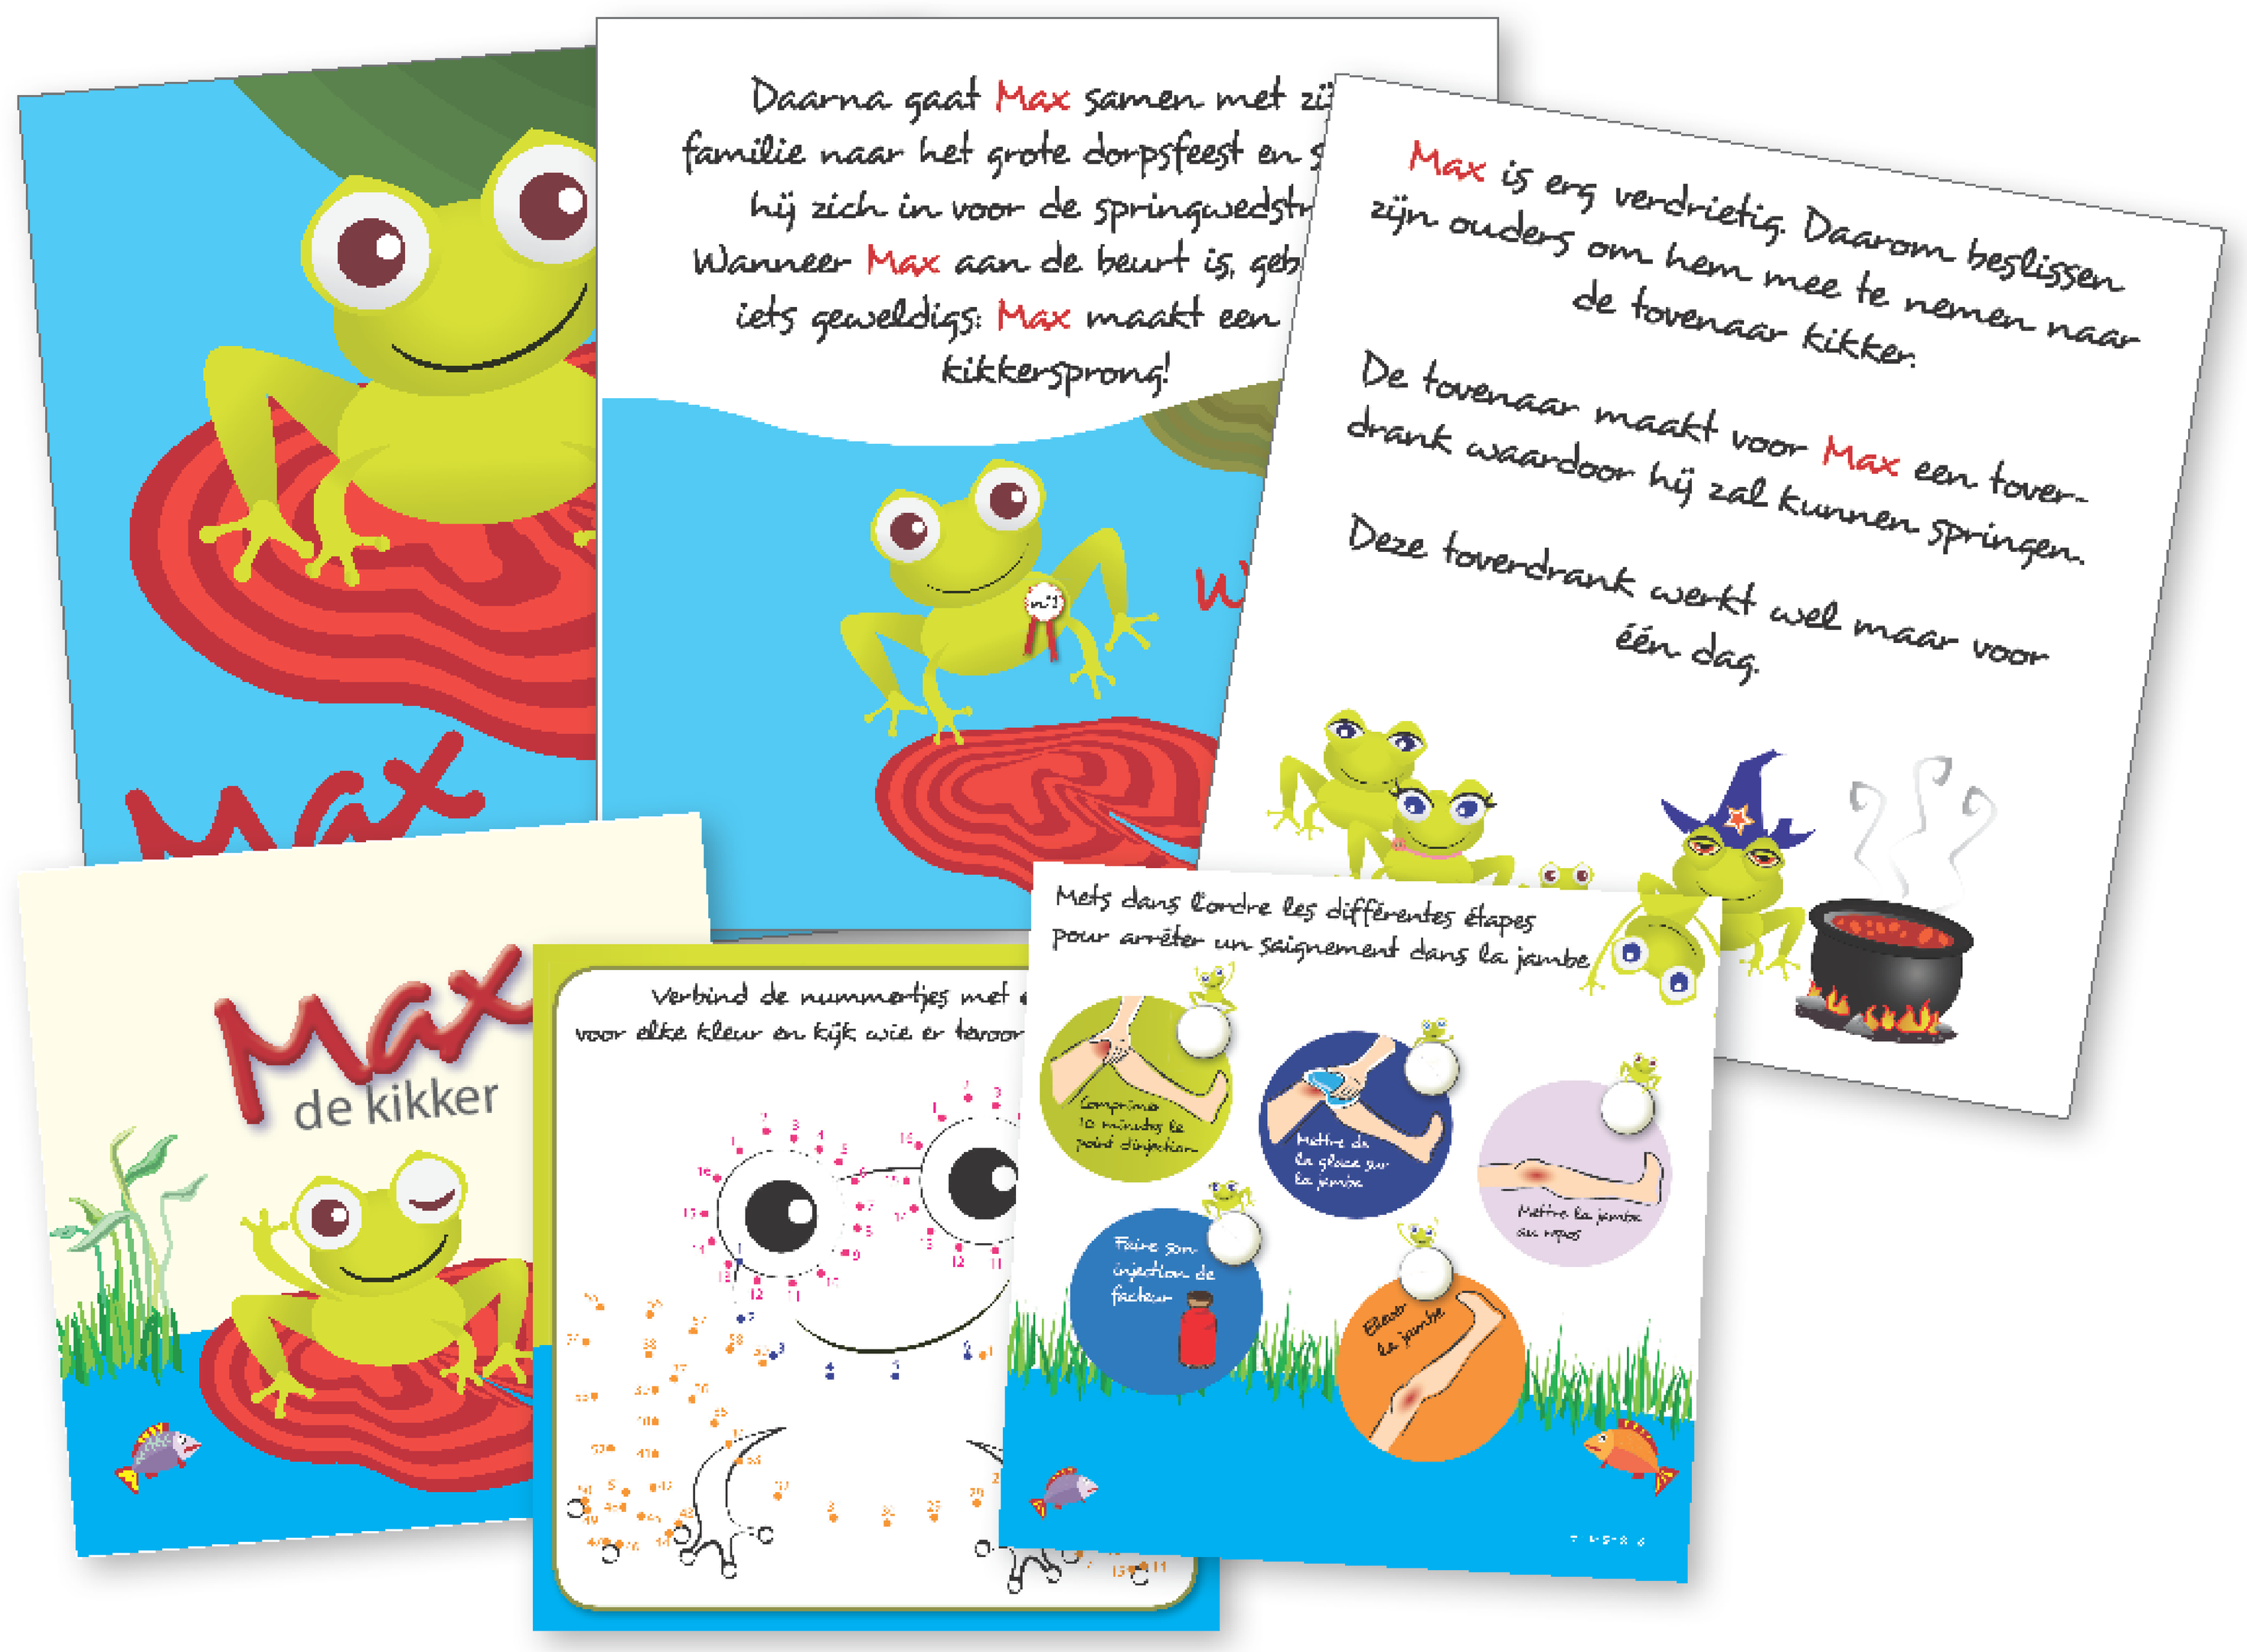
Max is erg verdrietig. Daarom beslissen zijn ouders om hem mee te nemen naar de tovenaar kikker.
De tovenaar maakt voor Max een toverdrank waardoor hij zal kunnen springen.
Deze toverdrank werkt wel maar voor één dag.
Max
de kikker
Mets dans l'ordre les différentes étapes pour arrêter un saignement dans la jambe

Material & Methods

"**Max the frog**" is an illustrated short story, intended for children aged between 4-6 years and on prophylaxis treatment. The story tries to make the link between Max's difficulty jumping and the bleeding problems of children with hemophilia. The story explains that Max, a frog who cannot jump, is not able to participate at the annual competition of the biggest jump. Thanks to his parents, he can drink a specific magic potion in order to jump again. This drink has a really bad taste but thanks to it he won the competition and can now play with his friends. Now he takes it regularly and is very happy!

The goal is to initiate a structured discussion with the child and check if he can make the connection between the story and his real life. So, we developed a questionnaire with several open-ended questions that allow the exchange between caregiver and child. The child can express themselves freely and give to caregiver the opportunity to assess the knowledge of the child and his experiences or concerns. In order to make a follow-up and reminder at home, we developed a play book with some games sometimes purely recreational and other educational.

Discussion & Conclusion

Max is an illustrated short story for young children, it is easy to understand because it uses language that is suited to their age. It is an excellent tool to use for communication with children. Using this tool we hope to make them understand more about hemophilia and, most importantly, the different aspects of their treatment. The goal of this tool is to obtain better compliance to treatment, reduce fear of injections and make children more familiar with hemophilia. The story is a nice and practical educational tool. This tool can be used in combination with other existing materials. It is an ideal tool to begin TEP with young patients.

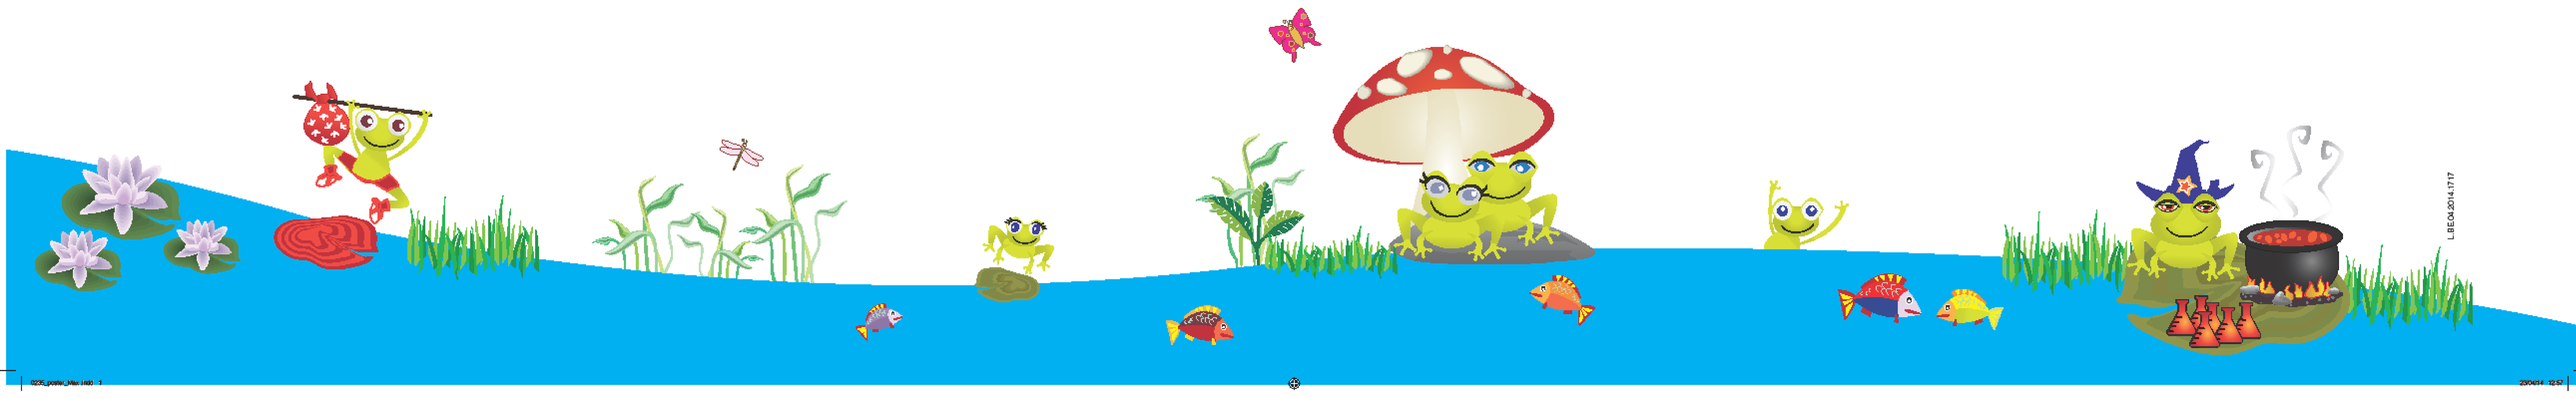

WFH2014 122--P-T

Educational Models

Franoise Delacolette

DOI: 10.3252/pso.eu.WFH2014.2014

Poster presented at: WFH 2014 World Congress

PosterSessionOnline.com