Physiological analysis of swallowing imparement caused by chronic obstructive pulmonary disease (COPD) in rats

Kouta Nagoya, Takanori Tsujimura, Midori Yoshihara, Makoto Inoue

Division of Dysphagia Rehabilitation, Niigata University Graduate School of Medical and Dental Sciences, Niigata, JAPAN

Introduction and Objective

Chronic obstructive pulmonary disease (COPD) is a respiratory illness, of which the number of patients is expected to increase with age. Dyspnea, cough and sputum are known as a main symptom of COPD, often accompanied by difficulty in swallow (ex; discoordination between respiration and swallowing, delayed swallowing reflex occurring, poor laryngeal elevation, poor UES opening). Although many clinical reports dealing with swallowing disorder related to COPD have been published, physiological mechanism of swallowing changes due to COPD still remains unclear. So, we analyzed how COPD contributes to swallowing impairment using COPD model rats.

Procedures

Animals: 60 Sprague Dawley male rats (300 - 350 g, Charles River Laboratories, Yokohama, Japan)
Introduction to COPD: The 28U/BW100 g of porcine pancreatic elastase (PPE) dissolved in 100 µl of sterile PBS was inhaled by intratracheal injection using a MicroSprayer drug delivery device. Three weeks later, rats were given intratracheal administration of 5 mg/mL lipopolysaccharide (LPS) dissolved in 100 µl of PBS.

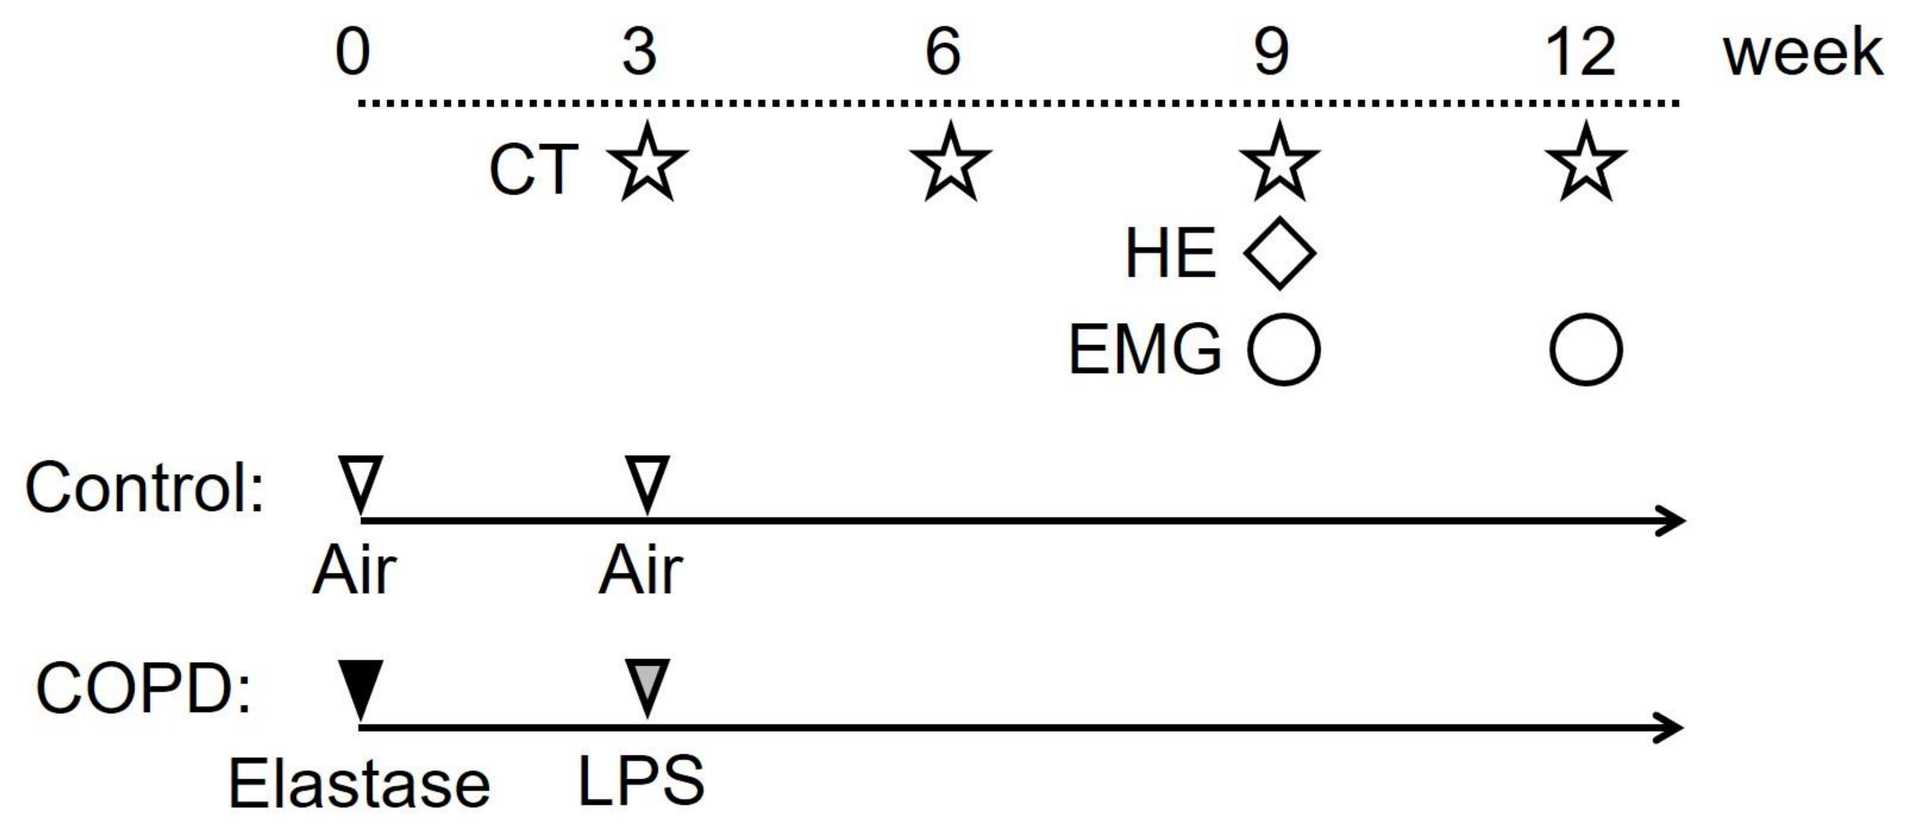
0
3
6
9
12
week
CT
HE
EMG
Control:
Air
Air
COPD:
Elastase
LPS

Flow chart of experiment to establish COPD model rats

At the point of 3, 6, 9 and 12 weeks after PPE administration, the lungs were analyzed using micro-CT (CosmoScan GX, RigakuCorporation) and HE staining.

EMG recording: To identify swallowing and respiration, electromyographic activity was recorded from digastric (Dig) and thyrohyoid (TH) muscles and diaphragm (Dia). All recordings were performed under the anesthetized condition with urethane (1.3 g/kg, ip).
Swallowing initiation: The superior laryngeal nerve (SLN) was electrically stimulated repetitively (0.2 ms pulse duration; 30 Hz). The stimulation threshold for evoking swallow was determined as the minimum stimulus intensity needed to evoke a swallow at least once during the SLN stimulation for 10 s. The current intensity was determined as 1.2 or 2 times the stimulation threshold.
Swallowing analysis: The muscle activity (duration, onset to peak, peak to offset) was analyzed by EMG.
Respiration analysis: The duty cycle was calculated as a ratio of the inspiratory duration to the total respiratory duration.

Results

1. The normalized BW of COPD model rats was significantly lower than that of control rats.

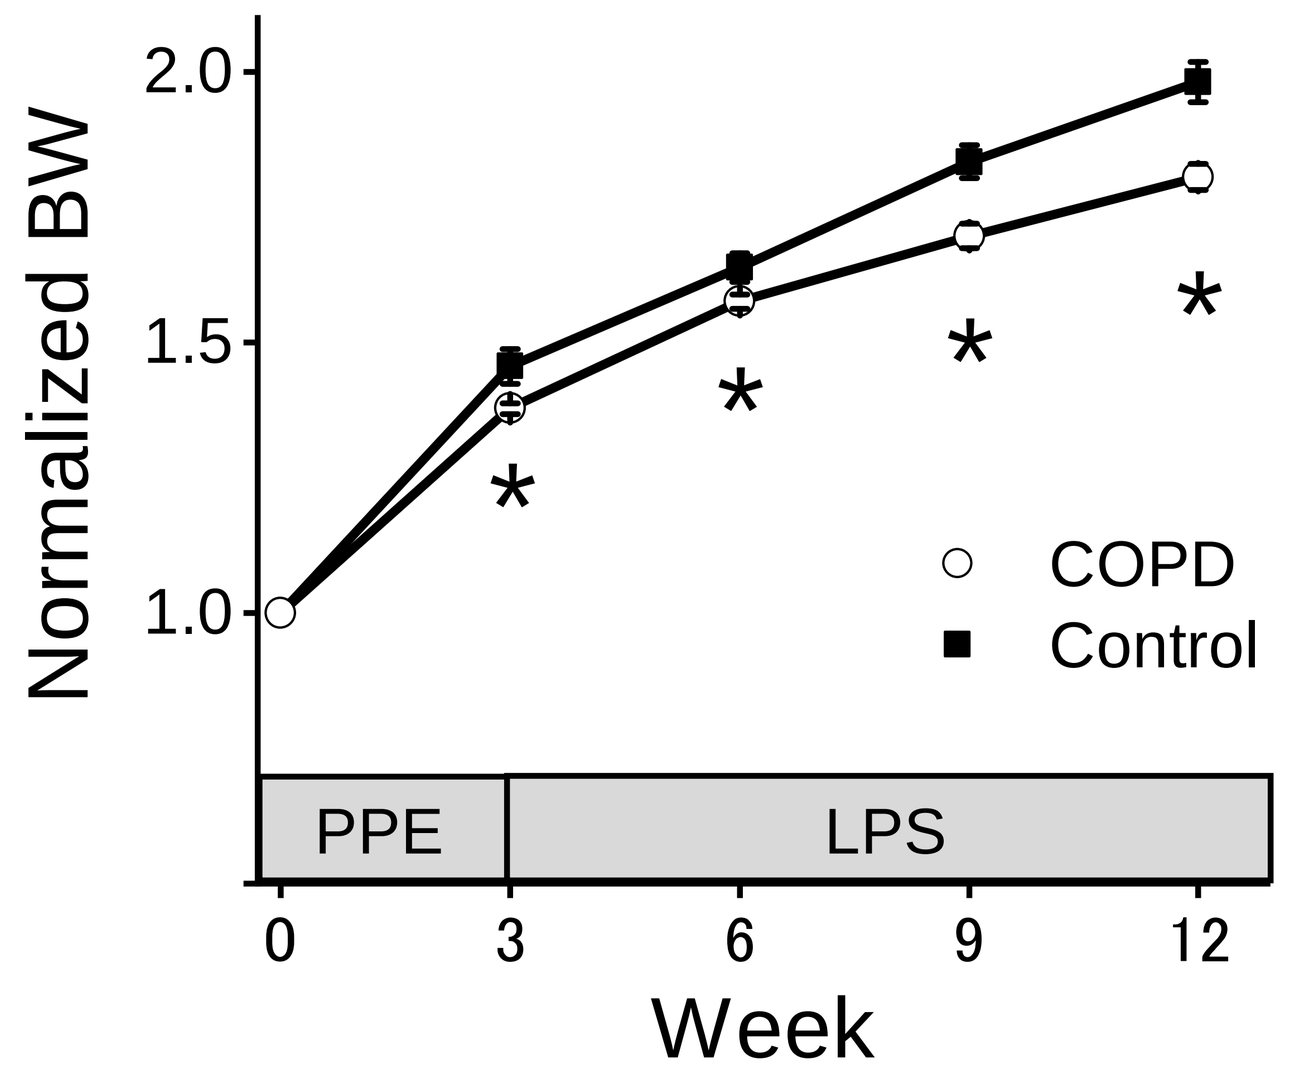
Normalized BW
2.0
1.5
1.0
COPD
Control
PPE
LPS
0
3
6
9
12
Week

The body weight (BW) of COPD model and control rats was measured every 3 weeks.
The normalized BW was a ratio compared with BW at PPE injection.
We confirmed that the normalized BW of COPD model rats was significantly lower compared with control rats.
*P<0.05 (Student`s t-test).

2. COPD condition was confirmed using micro-CT analysis and HE staining.

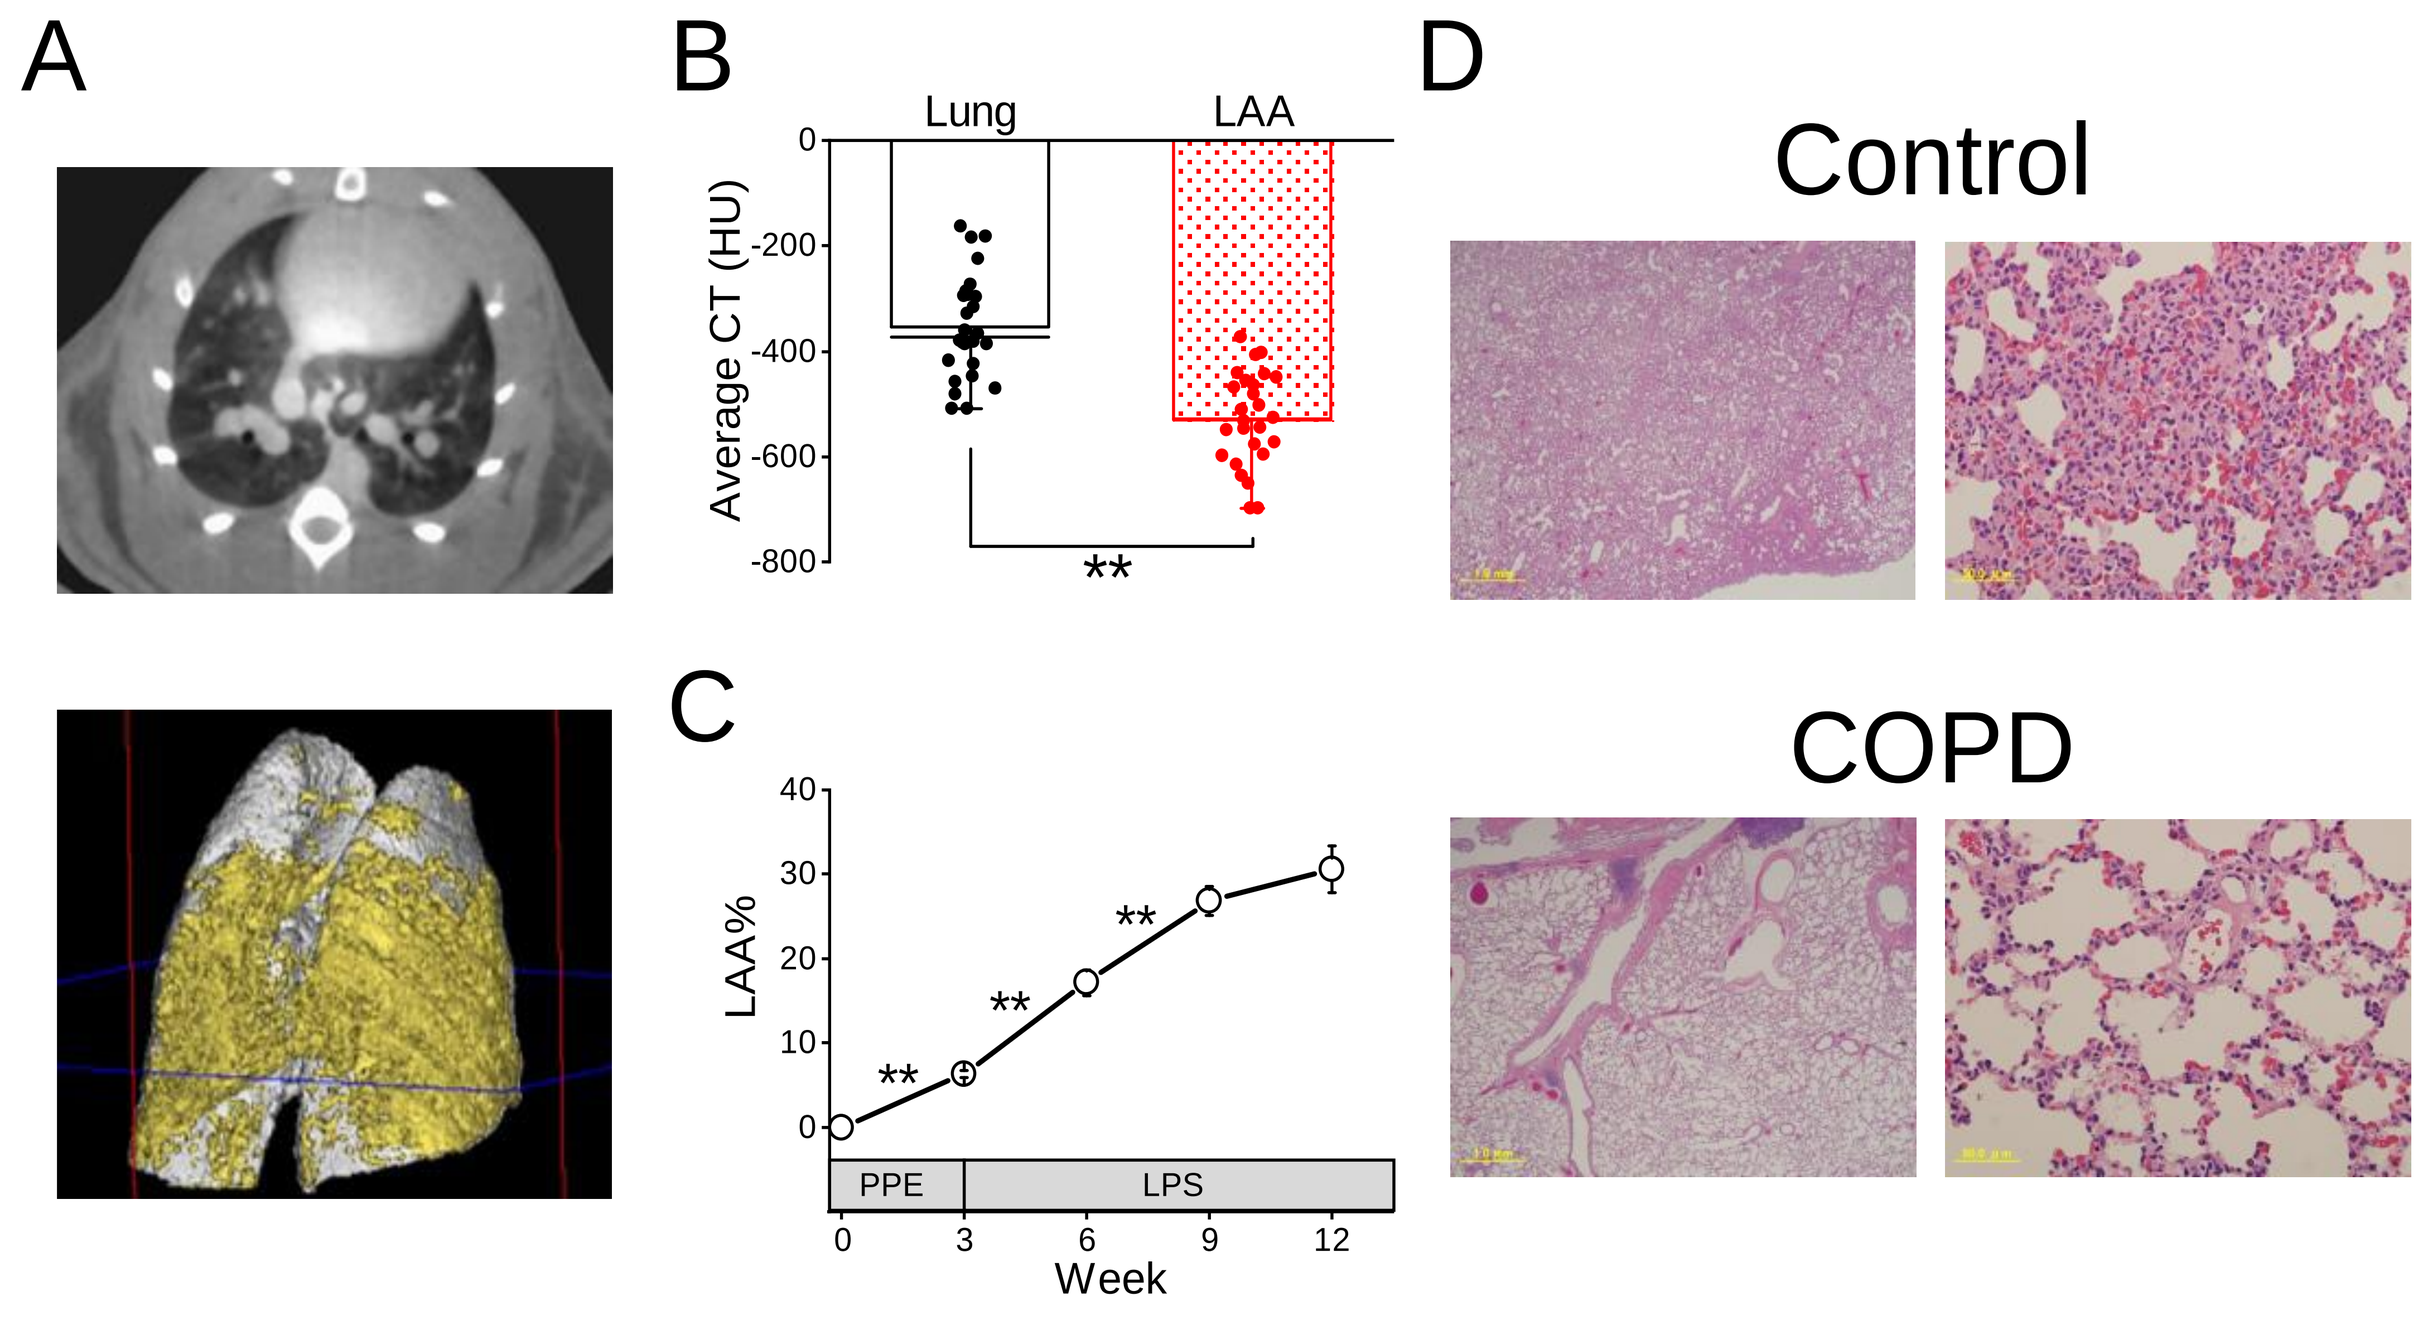
A
B
Lung
LAA
Average CT (HU)
C
LAA%
PPE
LPS
Week
D
Control
COPD

A, representative transverse micro-CT image (upper) and three-dimensional reconstructed image (lower, gray; whole lung, yellow; emphysema) of COPD model rat at 9th week after PPE administration.
B, the low-attenuation area (LAA) thresholds were determined in the range of -400 to -700 Hounsfield units in our study.
C, LAA% of COPD model rats was significantly increased up to 9th week.
D, HE staining images of control and COPD model rat at 9th week.
**P<0.01 (Student`s t-test).

3. The duty cycle of COPD model rats was significantly higher than that of control rats.

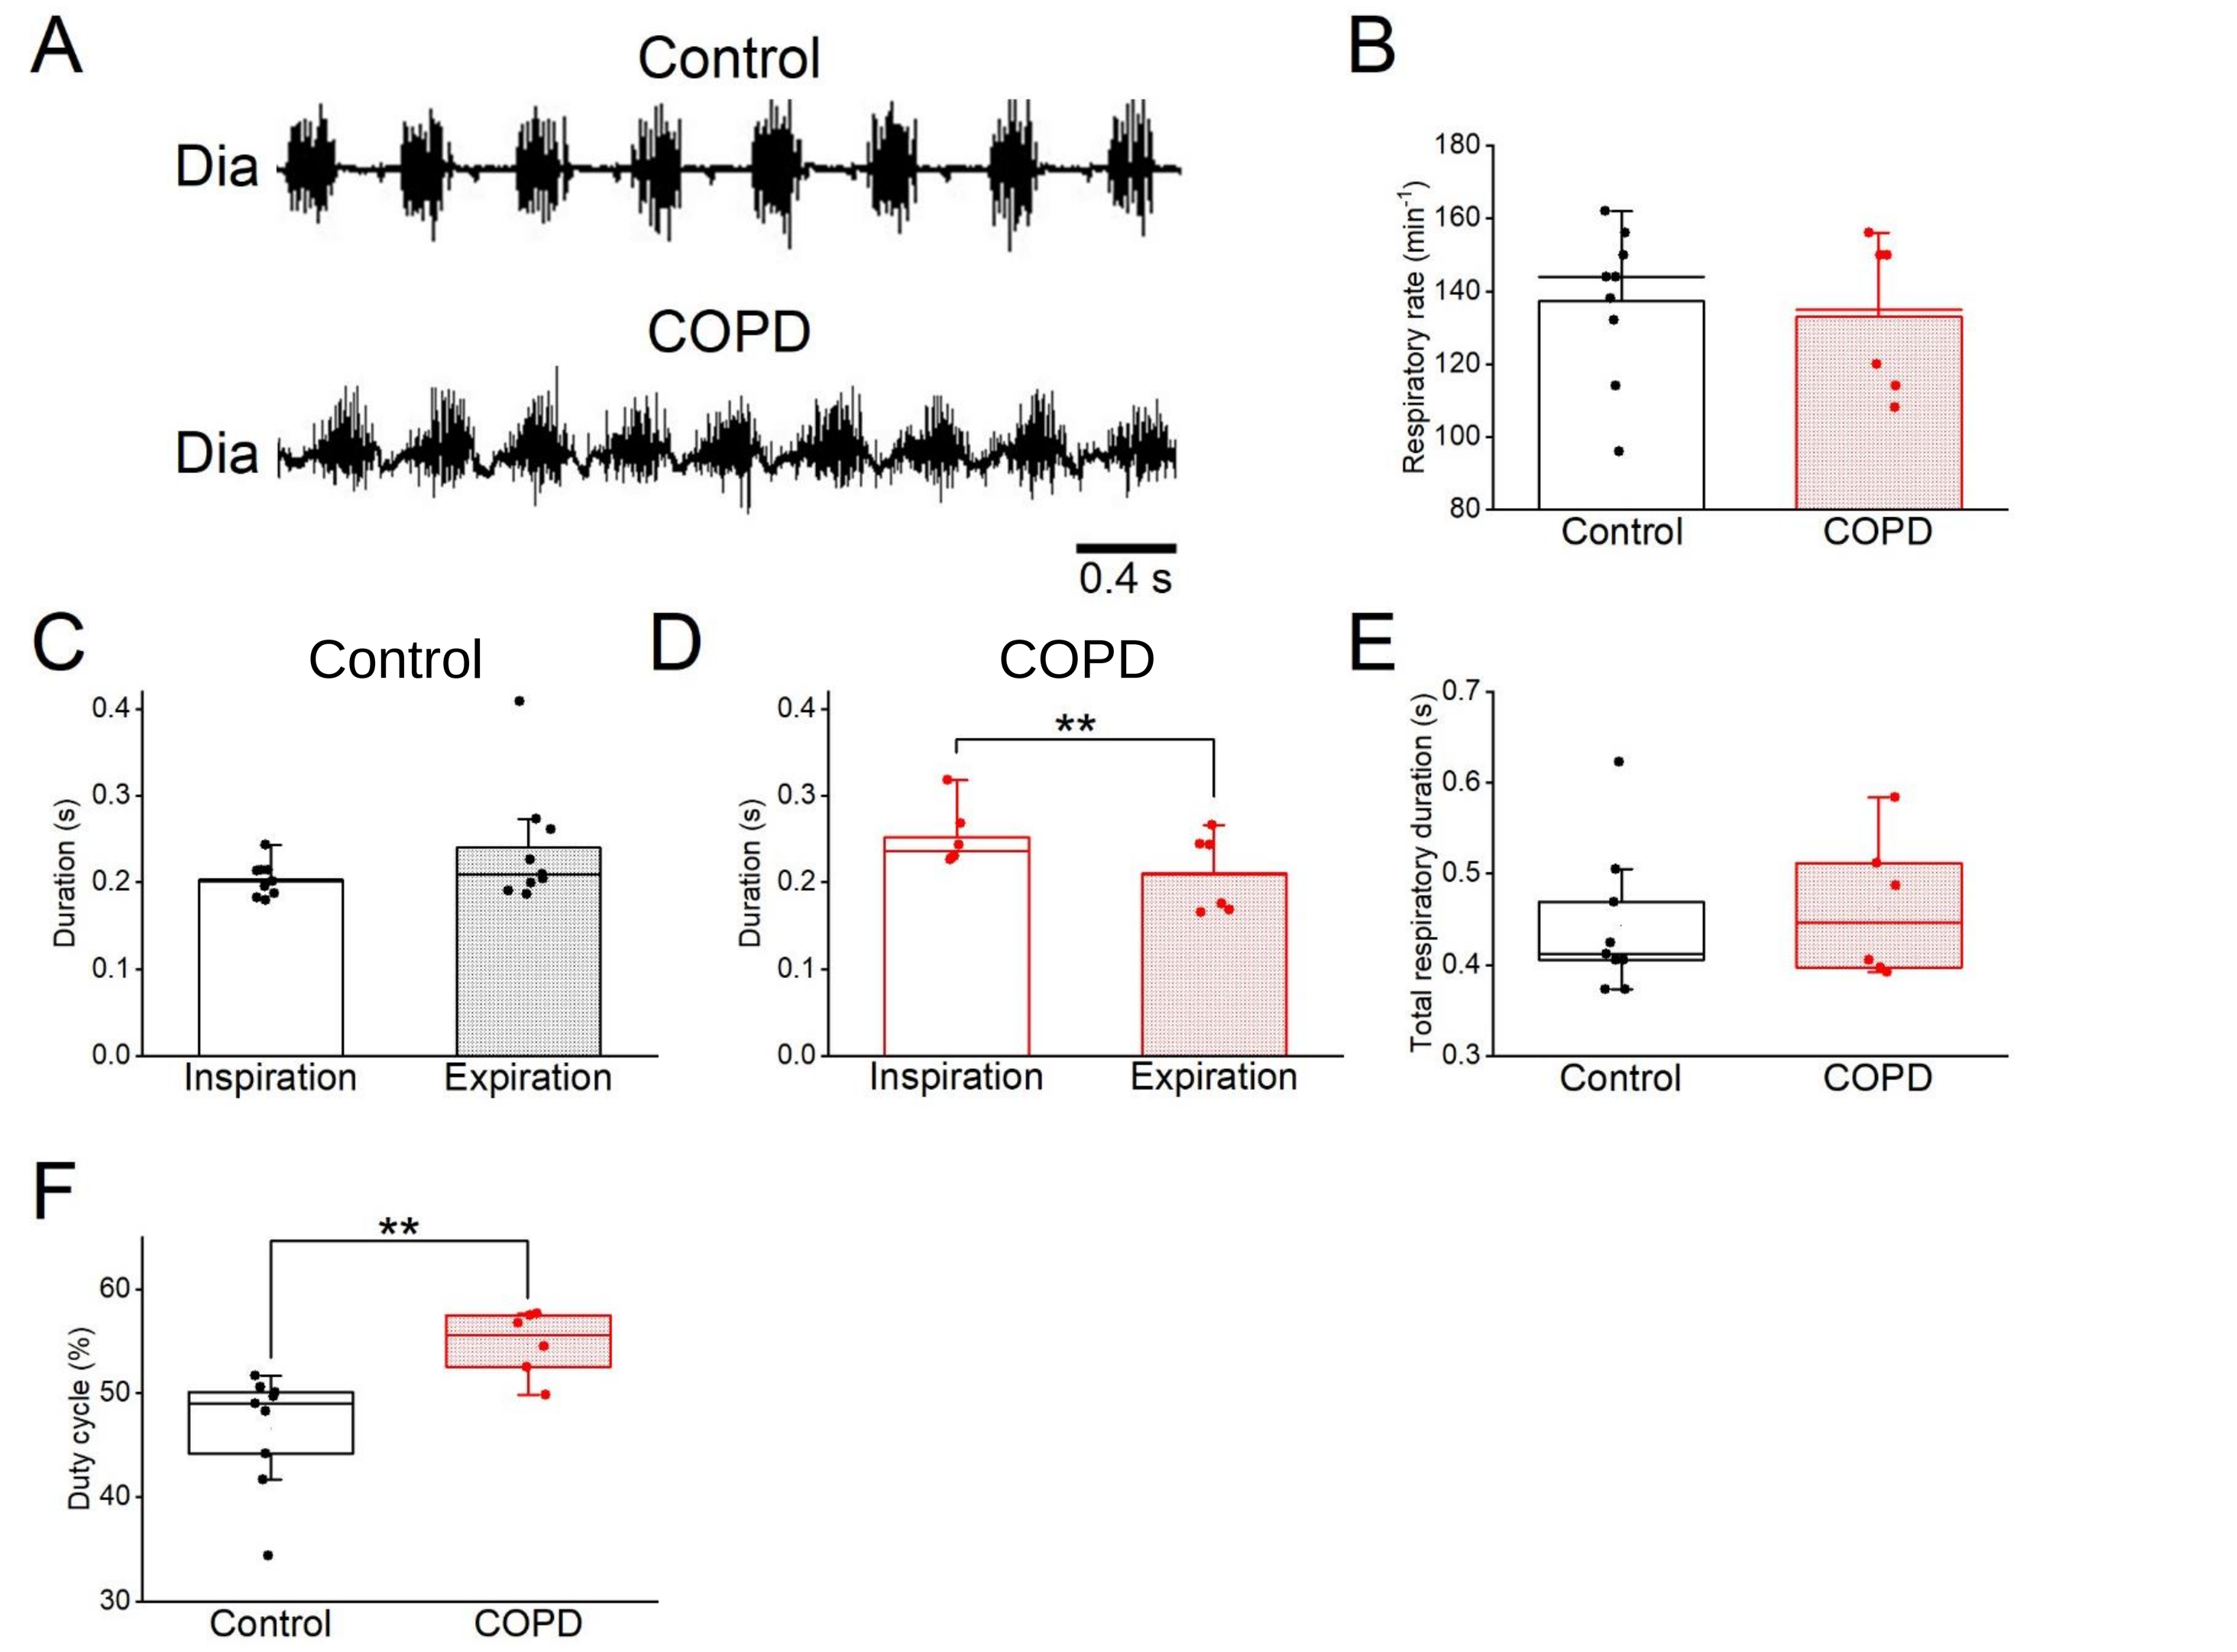
A
Control
Dia
COPD
Dia
0.4 s
B
Respiratory rate (min-1)
Control
COPD
C
Control
Duration (s)
Inspiration
Expiration
D
COPD
E
Total respiratory duration (s)
F
Duty cycle (%)

A, typical Dia EMG traces at eupnea of control (upper) and COPD model (lower) rats at 9th week.
B, the respiratory rate of COPD model rats did not show significant difference compared with control rats.
C and D, the mean duration of inspiration and expiration of control (C) and COPD model rats (D). The inspiratory duration was significantly longer than the expiratory duration in COPD model rats.
E, the total respiratory duration did not indicate significant difference between control and COPD model rats.
F, the duty cycle of COPD model rats was significantly higher than control rats.
**P<0.01 (Student`s t-test).

4.Occurrence frequency of swallowing during the inspiratory phase was higher in COPD model rats.

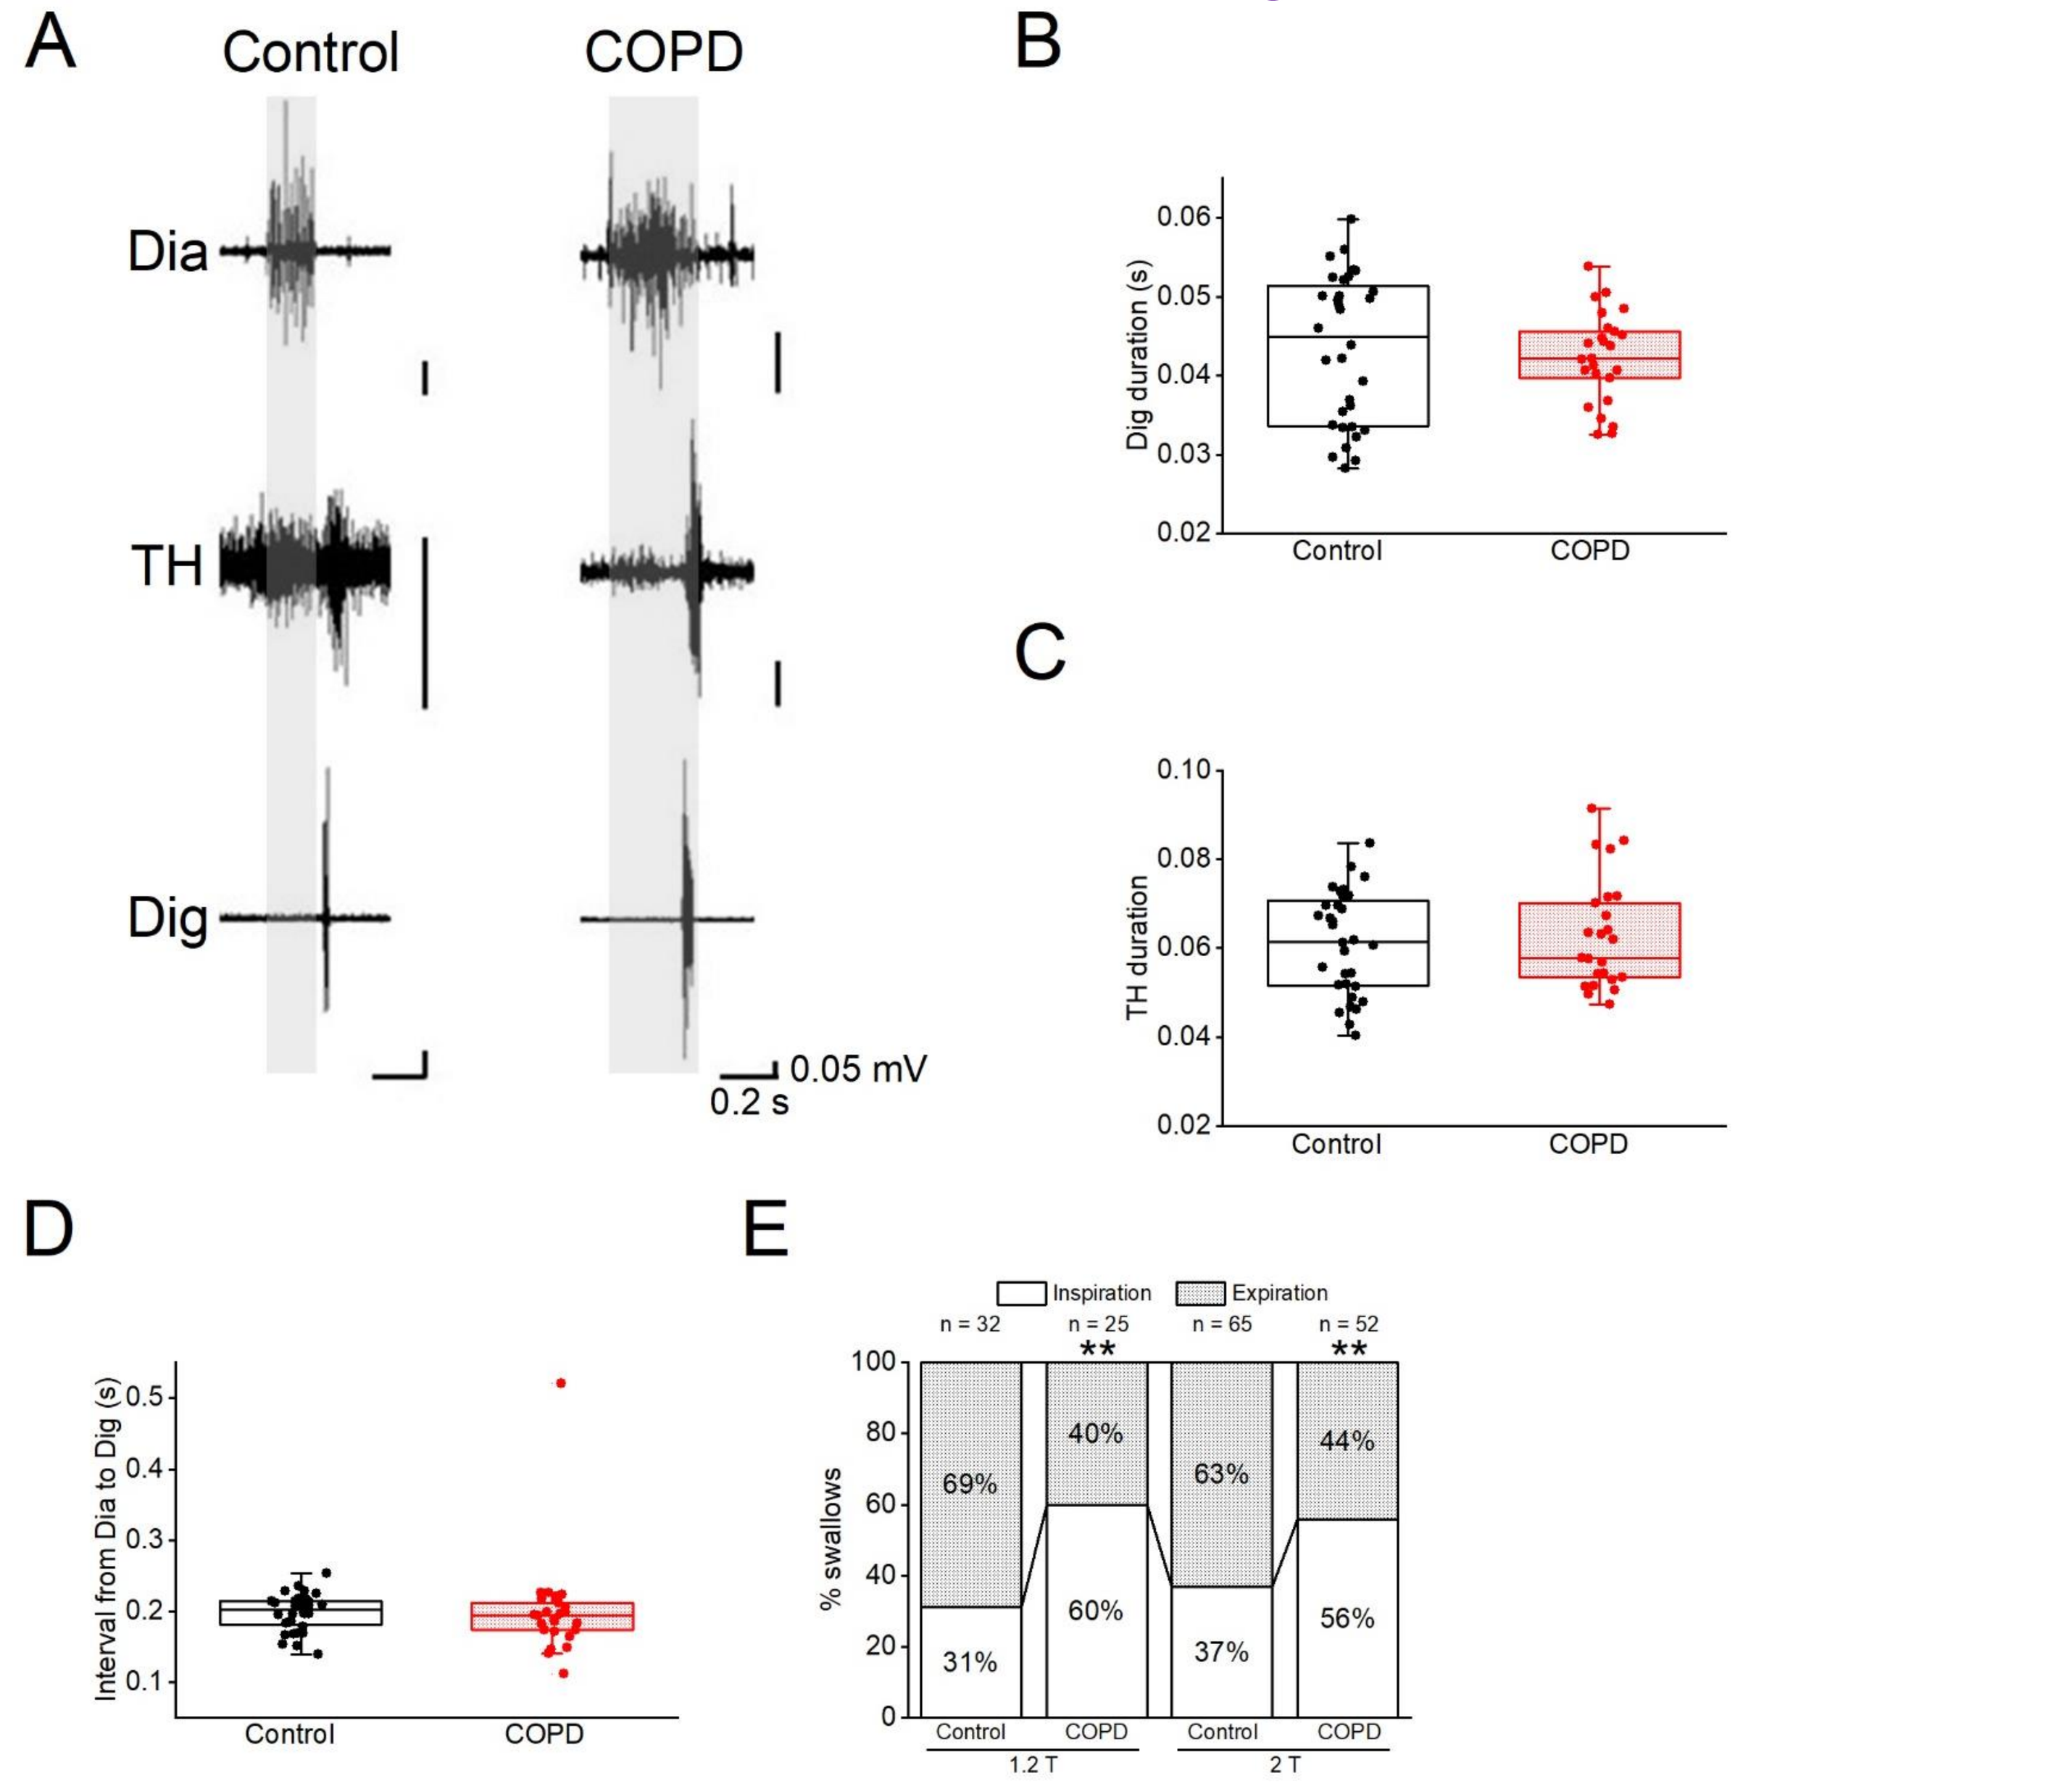
A
Control
COPD
Dia
TH
Dig
0.05 mV
0.2 s
B
Dig duration (s)
C
TH duration
D
Interval from Dia to Dig (s)
E
Inspiration
Expiration
n = 32
n = 25
n = 65
n = 52
% swallows
69%
31%
40%
60%
63%
37%
44%
56%
1.2 T
2 T

A, representative EMG traces at swallowing reflex of control (left) and COPD model (right) rats at 9th week. The shade area demonstrate the inspiratory phase.
B and C, the Dig (B) and TH duration (C) did not show significant changes between control and COPD model rats.
D, the interval from inspiration starting to Dig activity occurring did not show significant difference between control and COPD model rats.
E, stacked bar chart showing percentages of swallowing reflex occurrence in the inspiratory phase (white column) and the expiratory phase (gray column) in control and COPD model rats. The results were consistent between 1.2 and 2 times the stimulation threshold.
**P<0.01 (χ^2test).

5. The long term COPD affected the swallowing related muscle activity.

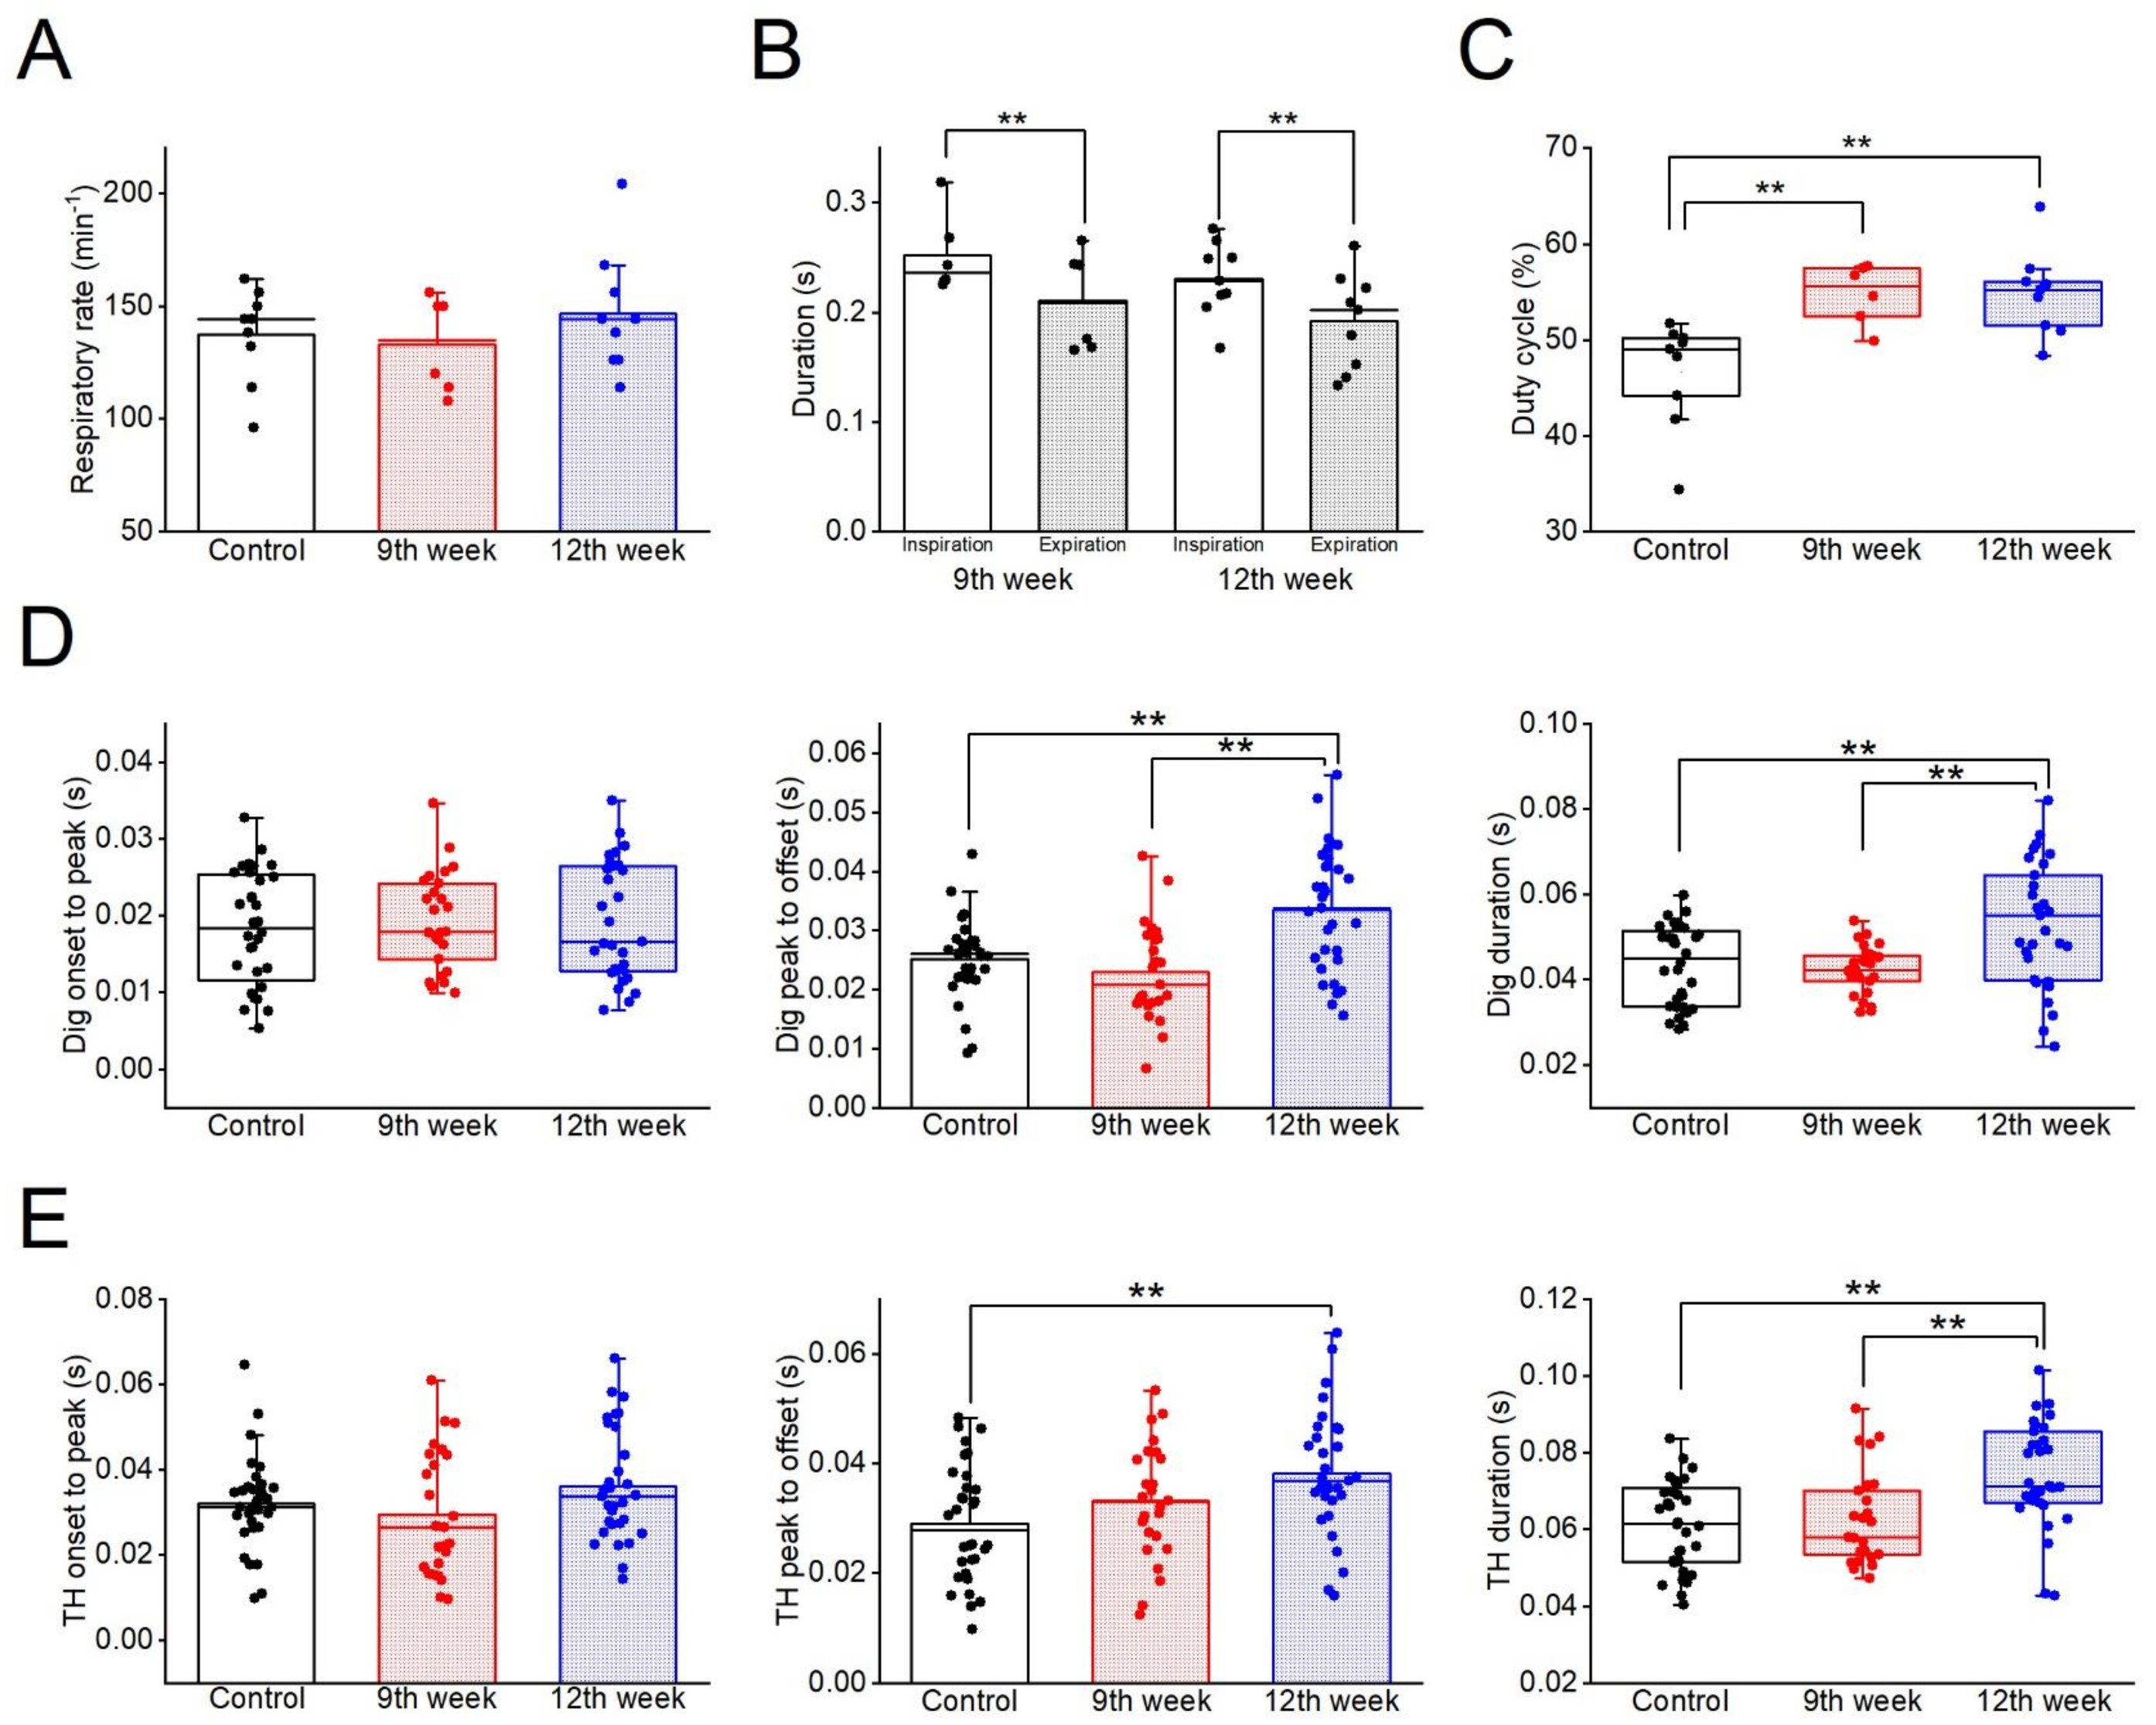
A
Respiratory rate (min-1)
B
Duration (s)
Inspiration
Expiration
9th week
12th week
C
Duty cycle (%)
D
Dig onset to peak (s)
Dig peak to offset (s)
Dig duration (s)
E
TH onset to peak (s)
TH peak to offset (s)
TH duration (s)
Control
9th week
12th week

A, B, C, the respiratory rate (A) and the duration of inspiration and expiration (B), the duty cycle (C) of COPD model rats at 12th week did not demonstrate significant differences compared with those at 9th week.
D, the peak to offset (middle) and the duration (right) of Dig of COPD model rats at 12th were significantly longer than those of control rats and COPD model rats at 9th week.
E, the peak to offset (middle) of TH of COPD model rats at 12th was significantly longer than that of control rats. The TH duration (right) of COPD model rats at 12th was significantly longer compared with control rats and COPD model rats at 9th week.
**P<0.01 (one-way ANOVA, Kruskal-wallis test).

6. Muscle atrophy and other pathological abnormal image were not confirmed in Dig and TH muscles of COPD model rats.

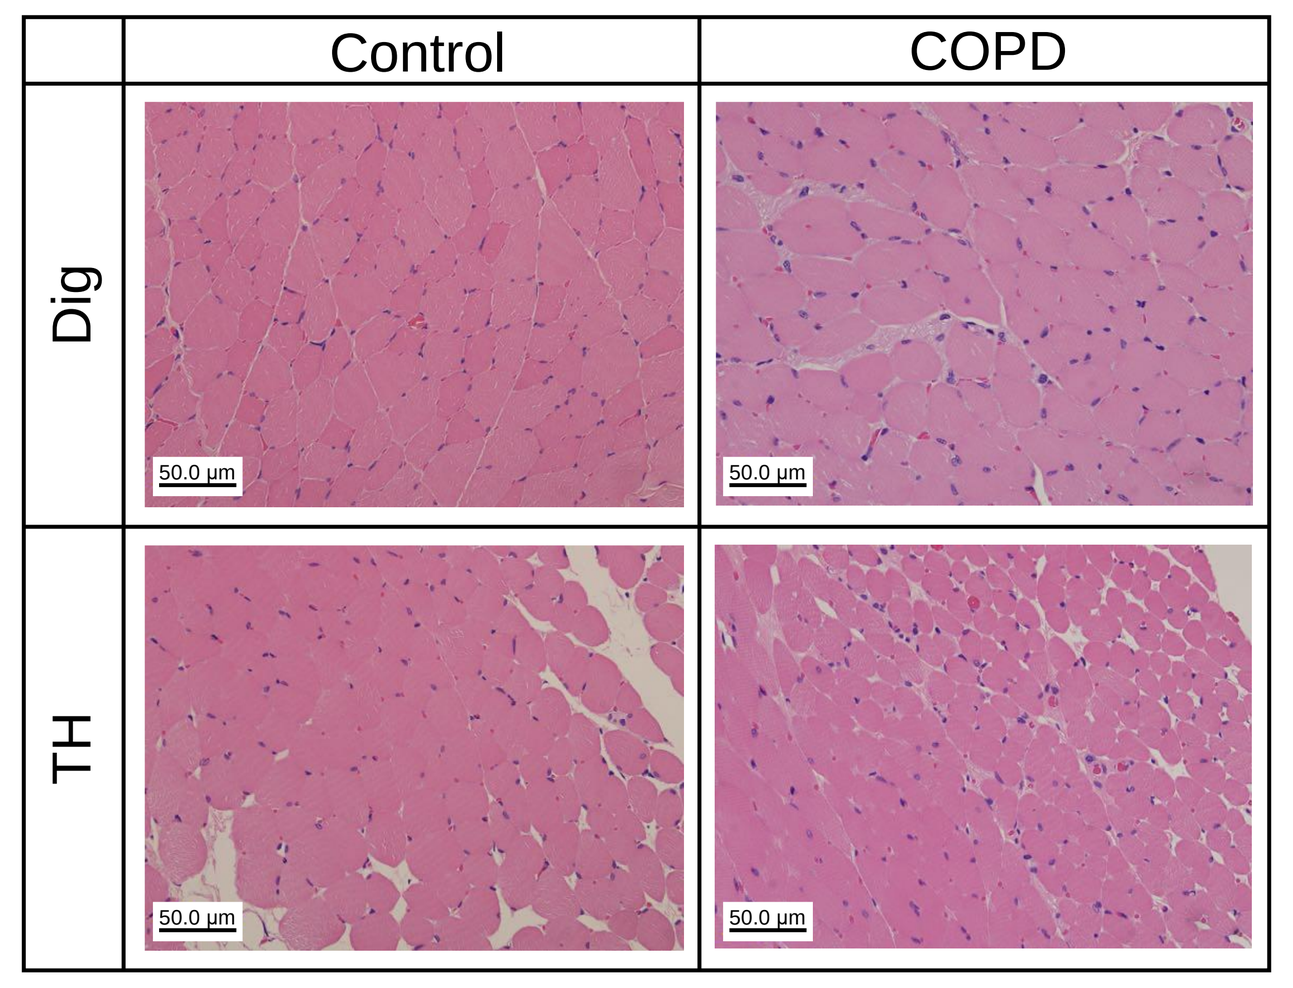
Control
COPD
Dig
TH
50.0 µm

Representative HE staining images of Dig (upper) and TH (lower) muscles in control (left) and COPD model (right) rats at 12th week.

There was no difference in the muscle cell number of Dig and TH between control and COPD model rats.

Summary and Conclusions

Our study suggested that the discoordination of respiration due to COPD increased the frequency of swallowing reflex during inspiration. Furthermore, the long term COPD affected the swallowing related muscle activity. Thus, these physiological changes may cause a high risk of aspiration in COPD patients.

This study was supported by a grant from GSK Japan Research Grant 2018. Authors have no conflict of interest in this study.

PosterSessionOnline.com
Poster presented at: DYSPHAGIA RESEARCH SOCIETY 29TH ANNUAL MEETING
Swallowing Physiology and Treatment
Nagoya Kouta
P-3487485
DRS2021