Ibrutinib at first relapse in mantle cell lymphoma: a United Kingdom real-world analysis of outcomes in 211 patients

Rory McCulloch[1], Simon Rule[2], Toby Eyre[3], Simon Bolam[4], Thomas Creasey[5], David Dutton[6], Safia Dawi[7], Matthew Wilson[8], Oliver Miles[9], Samuel Harrison[10], Harshita Goradia[11], Annabel McMillan[12], Graham McIlroy[13], Andrew Robinson[14] , Chibuzo Mowete[15], Michelle Furtado[16], David Tucker[16], Luke Attwell[17], Jamie Maddox[18], Amir Shenouda[19], Nimish Shah[20], Adam Bond[21], Phillipa Kelsey[22], Ahmed Abdulgawad[23], Jennifer Clay[24], Alexander Langridge[25], Deborah Turner[26] , Helen Parry[19], Richard Buka[27], Angharad Pryce[28], Anita Arasaretnam[29], John Willan[30], Sara Willoughby[31], Rebecca Walker[32], Gavin Campbell[33], Neil Phillips[10], Helen McCarthy[17], Rosalynd Johnston[29], Jonathan Lambert[12], Wendy Osborne[5], Pam McKay[8], Mark Bishton[11], David Lewis[1], Nicola Crosbie[1]

[1]University Hospitals Plymouth; [2]University of Plymouth; [3]Oxford University Hospitals; [4]Musgrove Park Hospital, Taunton; [5]Newcastle upon Tyne Hospitals; [6]University Hospital Southampton; [7]Hull Teaching Hospitals; [8]Beatson West of Scotland Cancer Centre, Glasgow; [9]Gloucestershire Hospitals; [10]Royal Stoke University Hospital; [11]Nottingham University Hospitals; [12]University College London Hospitals; [13]University Hospitals Coventry & Warwickshire; [14]Cambridge University Hospitals; [15]Kent & Canterbury Hospital; [16]Royal Cornwall Hospital; [17]Royal Bournemouth Hospital; [18]South Tees Hospitals; [19]University Hospitals Birmingham; [20]Norfolk & Norwich University Hospital; [21]Bristol Royal Infirmary; [22]Sheffield Teaching Hospitals; [23]Blackpool Victoria Hospital; [24]Leeds Teaching Hospitals; [25]Sunderland Royal Hospital; [26]Torbay Hospital; [27]Birmingham City Hospital; [28]Hammersmith Hospital; [29]Brighton & Sussex University Hospital; [30]Wexham Park Hospital; [31]Hereford Hospital; [32]QE Gateshead; [33]Colchester Hospital.

Background

The oral BTK inhibitor ibrutinib has transformed the clinical approach to relapsed mantle cell lymphoma (MCL). Clinical trials show those receiving ibrutinib at first relapse appear to obtain greatest survival benefit[1]. However, the general applicability of clinical trial findings is not established and concerns regarding ibrutinib tolerability persist, especially in non-trial populations enriched for older patients with multiple co-morbidities. In the United Kingdom, ibrutinib is funded as standard of care at first relapse. This study has analysed the clinical effectiveness and tolerability of this approach in a real world population and provides insights on post-ibrutinib outcomes.

Method

Hospitals across the UK were invited to contribute retrospective, anonymised data on all patients with MCL receiving ibrutinib at first relapse. For inclusion patients required confirmed diagnosis of relapsed or refractory MCL and had received only one prior line of systemic therapy prior to starting ibrutinib monotherapy. All patients commenced ibrutinib after January 2015, when ibrutinib first became available on the NHS. Patients started ibrutinib prior to July 2019. The database was locked in June 2020 for analysis.

Baseline characteristics

Characteristic	
Median age, years (range)	73 (33-96)
Male (*n*=211)	147 (70%)
Performance status (*n*=193)	
ECOG 0-1	147 (76%)
ECOG 2	36 (19%)
ECOG 3-4	10 (5%)
Lactate dehydrogenase ratio (*n*=147)	
<1.0	75 (51%)
≥1.0	72 (49%)
White cell count (n=197)	
<10 x 10^9/L	137 (70%)
≥10 x 10^9/L	60 (30%)
Simplified MIPI group (*n*=142)	
Low risk	19 (13%)
Intermediate risk	58 (41%)
High risk	65 (46%)
CNS disease (*n*=211)	
Absent	203 (96%)
Present	8 (4%)
Histology* (*n*=205)	
Non-blastoid	176 (86%)
Blastoid	29 (14%)
Stage* (*n*=208)	
I-II	14 (7%)
III-IV	194 (93%)
Frontline therapy (*n*=211)	
R-CHOP	66 (31%)
High dose cytarabine based regimen	60 (28%)
R-Bendamustine	45 (21%)
Chlorambucil +/-R	15 (7%)
Fludarabine, cyclophosphamide +/-R	9 (4%)
VR-CAP	5 (2%)
R-miniCHOP	4 (2%)
Other¥	7 (3%)
Stem cell transplant consolidation	
Autologous HSCT	50 (24%)
Allogeneic HSCT	3 (1%)

Outcomes for patients progressing on ibrutinib

100 patients discontinued ibrutinib due to progressive disease. In these patients median PFS with ibrutinib was 6.7 months (95% CI 4.9-8.4). Of 98 pts with available data, 56 pts (57%) received no additional systemic therapy after ibrutinib cessation and 42 pts (43%) received at least one additional therapy. At data lock 81 pts had died with median follow up from discontinuation 13 months for survivors. Median post-ibrutinib OS was 1.4 months (95% CI 0.6-2.2). Survival was significantly improved in those receiving further therapy (median post-ibrutinib OS 11.6 months, 95% CI 6.8-16.5, vs. 0.4 months, 95% CI 0.2-0.5). 23 patients of those receiving further therapy (55%) received R-BAC (rituximab, bendamustine, cytarabine), these pts displayed a strong trend to improved post-ibrutinib survival compared to patients receiving other therapies (median post-ibrutinib OS 14.0 months, 95% CI 8.1-19.8, vs. 3.6 months, 95% CI 2.6-4.5, P=0.06).

Fig. 2: **Post-ibrutinib OS for patients progressing on ibrutinib**

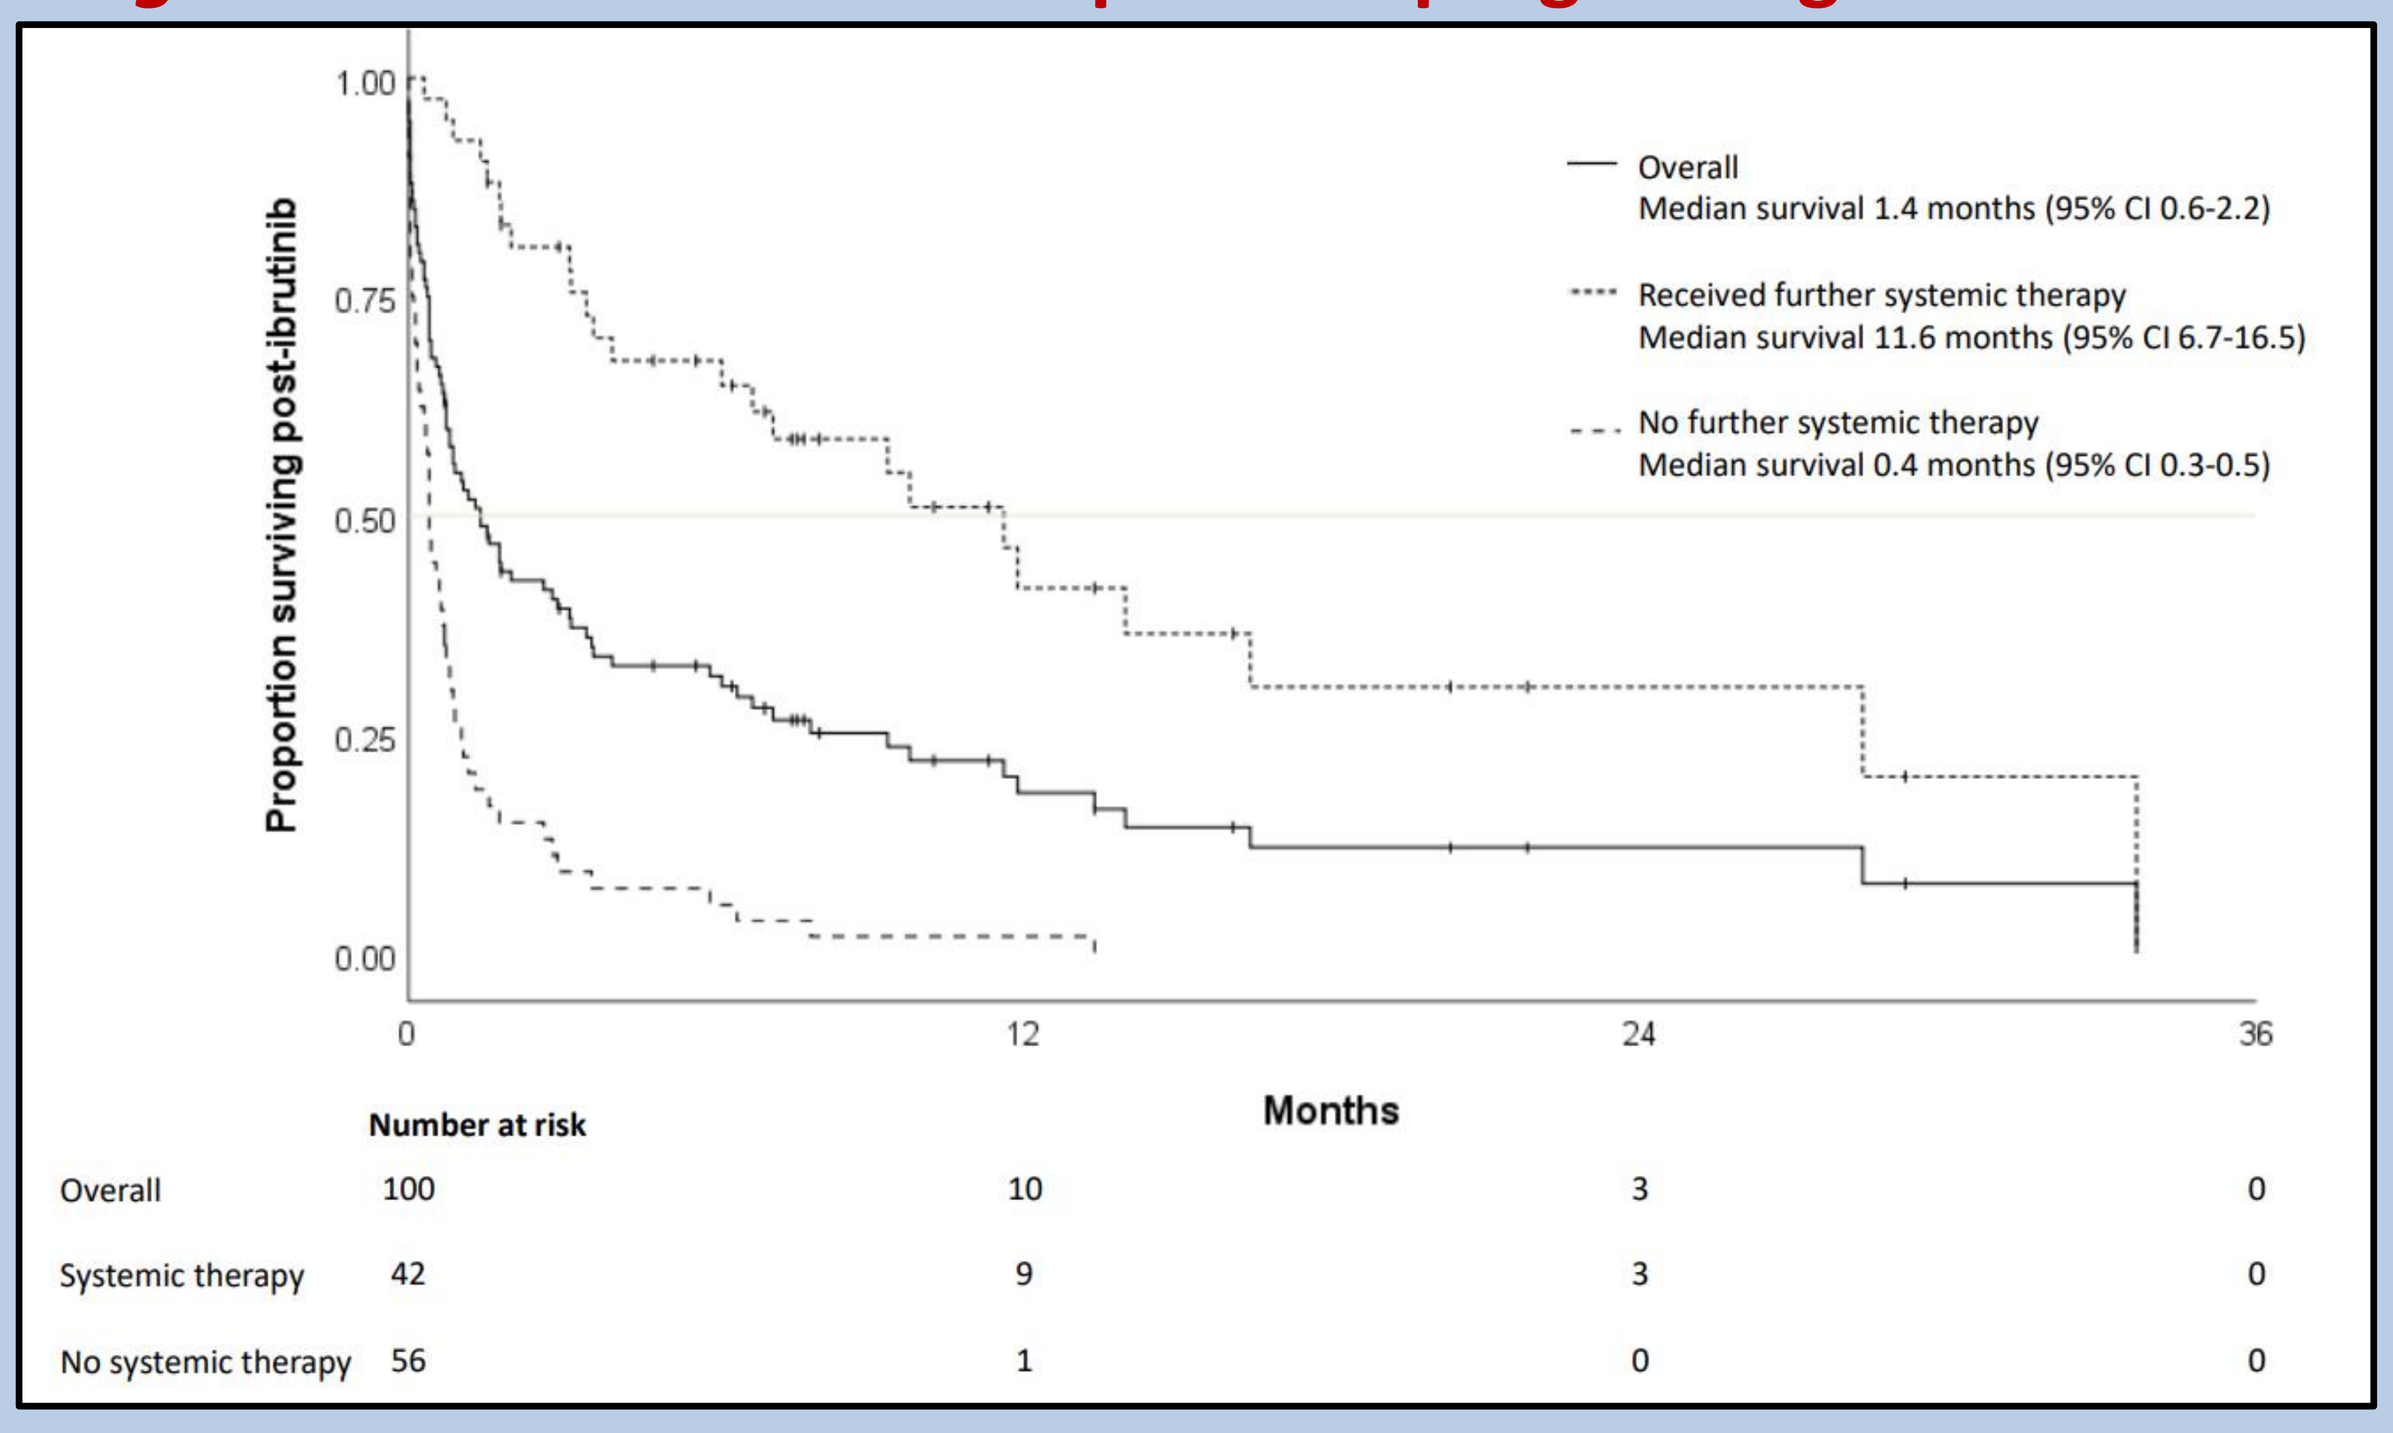
Proportion surviving post-ibrutinib
Overall
Median survival 1.4 months (95% CI 0.6-2.2)
Received further systemic therapy
Median survival 11.6 months (95% CI 6.7-16.5)
No further systemic therapy
Median survival 0.4 months (95% CI 0.3-0.5)
Months
Number at risk
Overall 100 10 3 0
Systemic therapy 42 9 3 0
No systemic therapy 56 1 0 0

Results

211 patients from 38 hospitals across the UK were included in the analysis. Median age at start of ibrutinib was 73 years (range 33 to 96 years). Eight patients had CNS involvement at start of ibrutinib. Median progression free survival (PFS) to frontline therapy was 21.4 months (95% CI 15.5 to 27.4). The overall response rate (ORR) to ibrutinib in evaluated patients was 69% with complete response (CR/CRu) 27% (49/179) and partial response (PR) 42%. At data-lock median follow-up of survivors by reverse censoring was 24 months (range 9-61 months), 118 patients had died, 59 remained on ibrutinib and 32 were alive having stopped ibrutinib (progressive disease [*n*=17], allogeneic (allo) HSCT [*n*=10], drug toxicity [*n*=4], patient choice [*n*=1]). The median PFS was 17.8 months (95% CI 13.1-22.2) and the median OS was 23.9 months (95% CI 15.0-32.8) (Fig. 1). PFS with ibrutinib exceeded PFS with frontline therapy in 40% of evaluable patients.

Fig. 1. Kaplan Meier survival analysis: Progression free survival with ibrutinib compared to frontline therapy (panel A); Overall survival from start of ibrutinib (panel B); PFS according to patient age, age <75 versus age≥75 (panel C) and PFS according to ECOG performance status at start of ibrutinib (panel D).

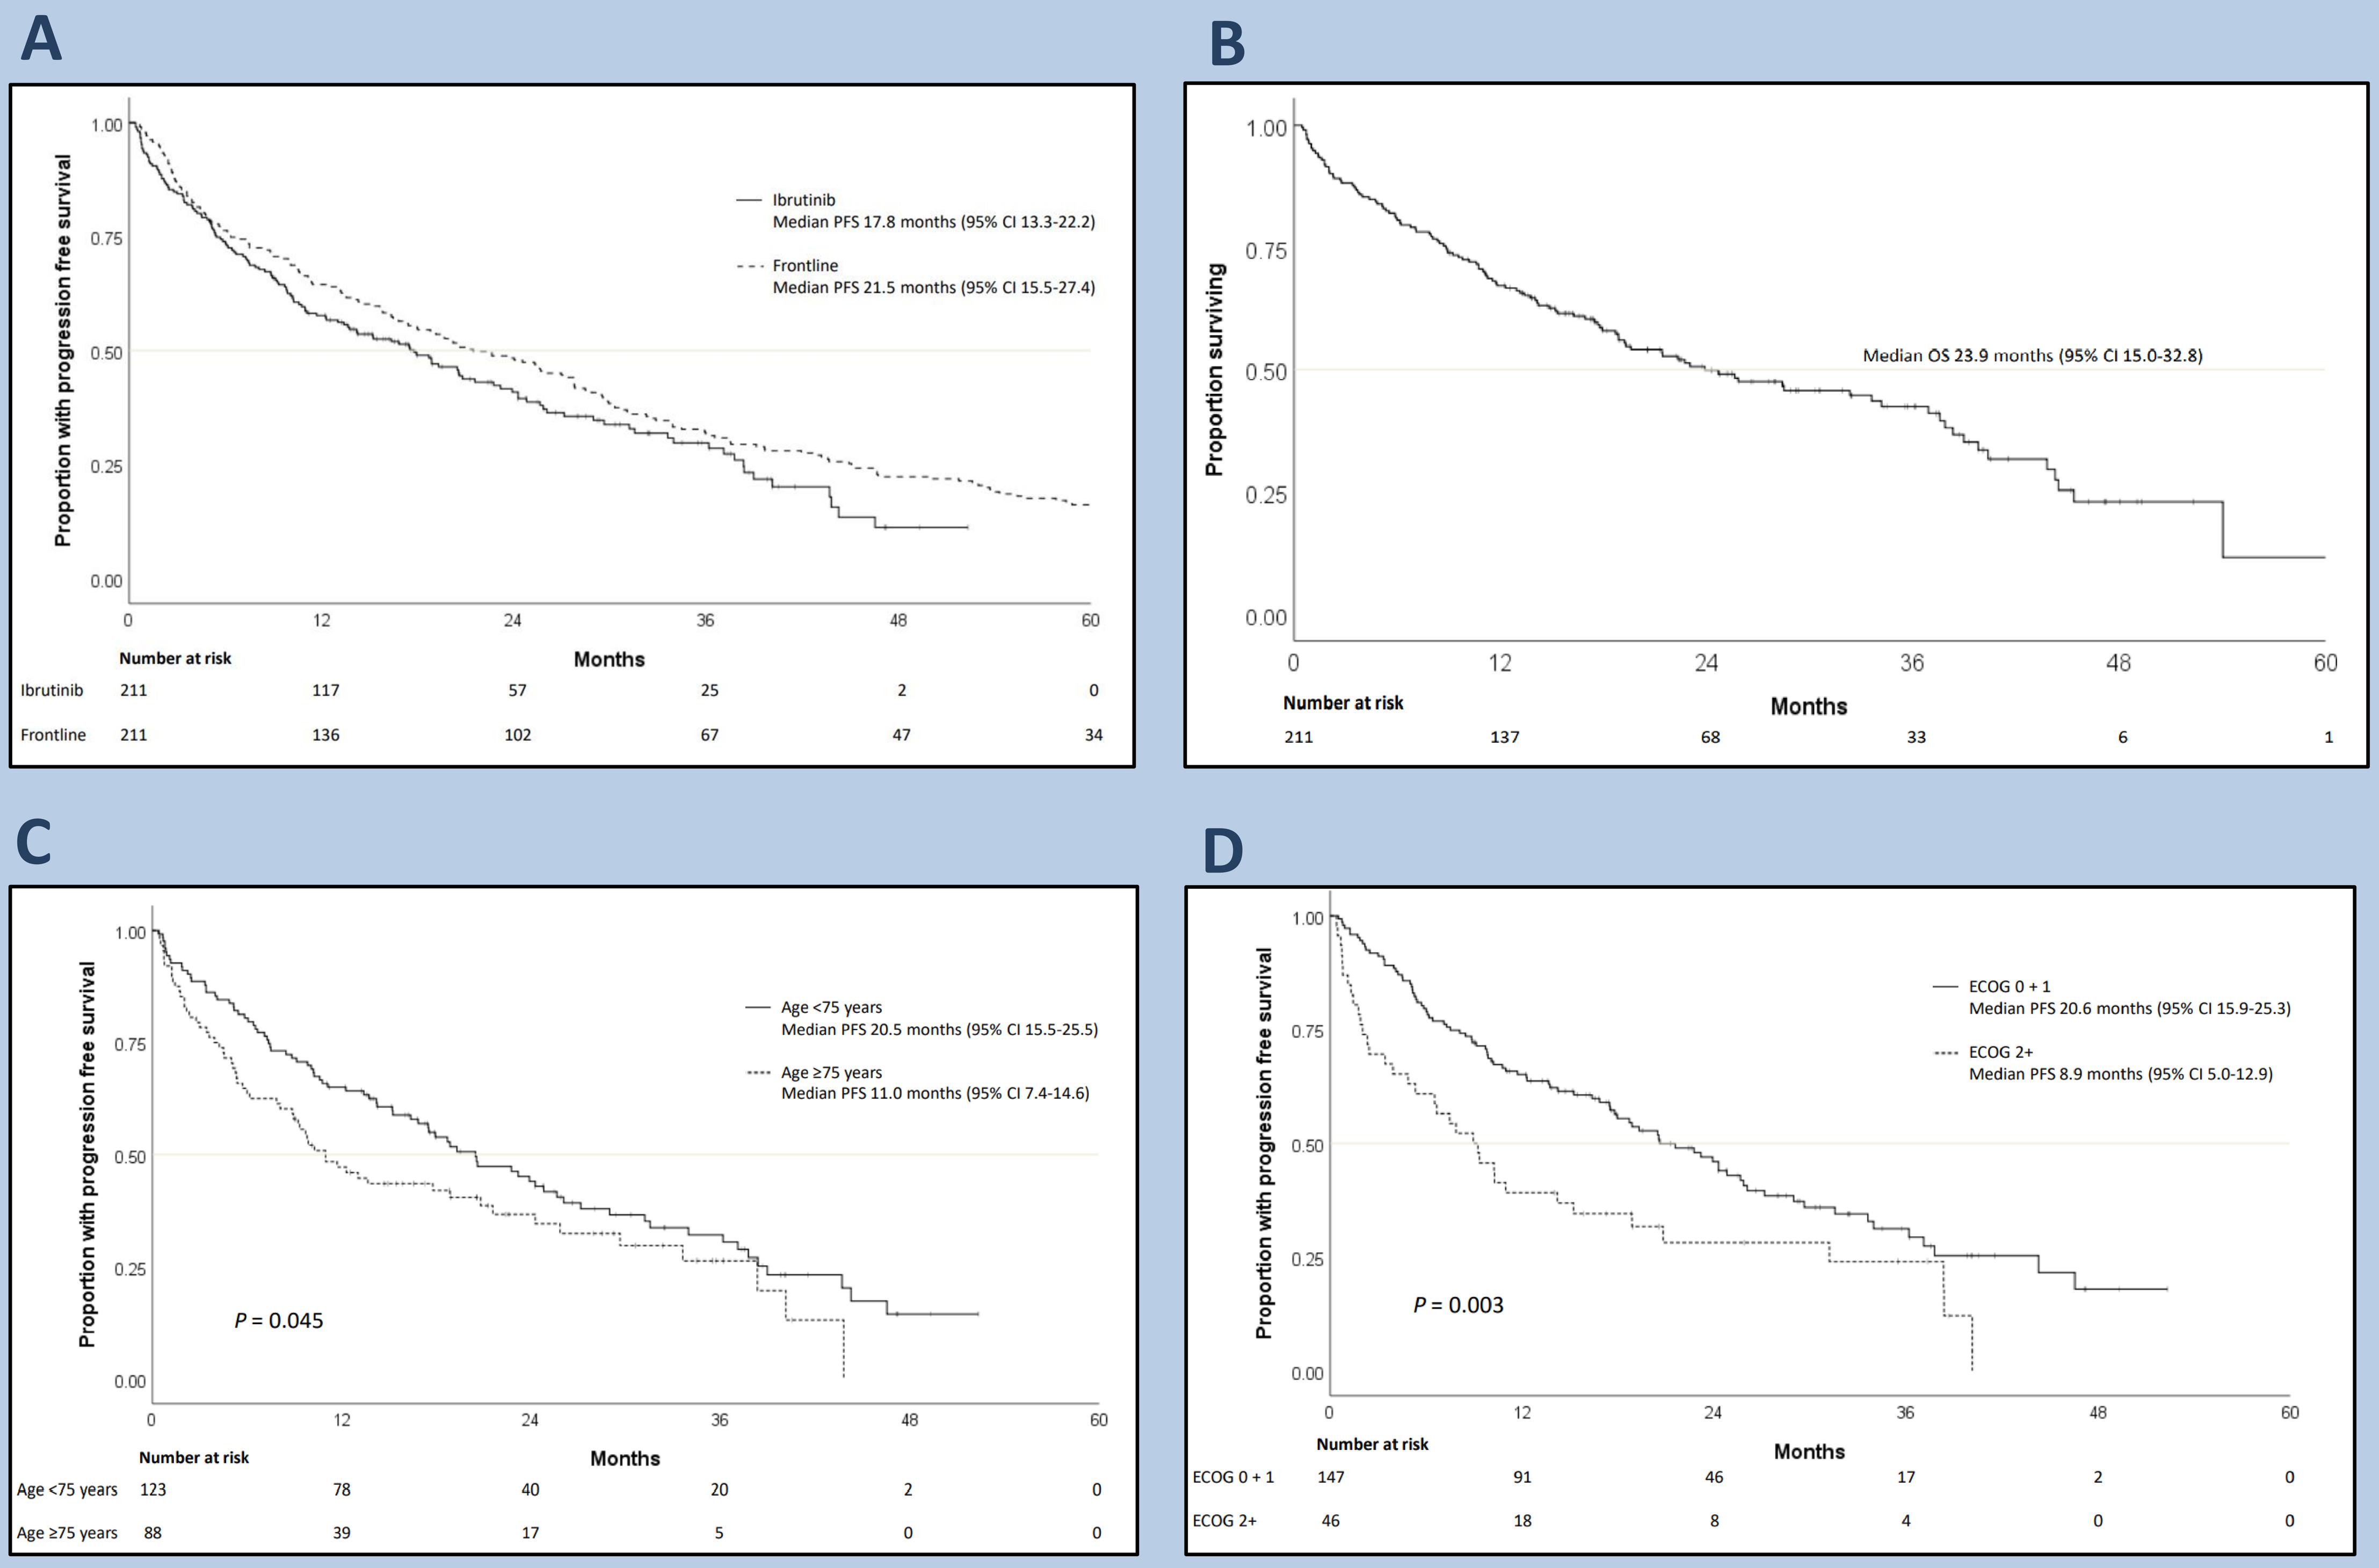
A
Proportion with progression free survival
Ibrutinib
Median PFS 17.8 months (95% CI 13.3-22.2)
Frontline
Median PFS 21.5 months (95% CI 15.5-27.4)
Months
Number at risk
Ibrutinib 211 117 57 25 2 0
Frontline 211 136 102 67 47 34
B
Proportion surviving
Median OS 23.9 months (95% CI 15.0-32.8)
Months
Number at risk
211 137 68 33 6 1
C
Proportion with progression free survival
Age <75 years
Median PFS 20.5 months (95% CI 15.5-25.5)
Age ≥75 years
Median PFS 11.0 months (95% CI 7.4-14.6)
P = 0.045
Months
Number at risk
Age <75 years 123 78 40 20 2 0
Age ≥75 years 88 39 17 5 0 0
D
Proportion with progression free survival
ECOG 0 + 1
Median PFS 20.6 months (95% CI 15.9-25.3)
ECOG 2+
Median PFS 8.9 months (95% CI 5.0-12.9)
P = 0.003
Months
Number at risk
ECOG 0 + 1 147 91 46 17 2 0
ECOG 2+ 46 18 8 4 0 0

Reasons for stopping ibrutinib

Indication	Number (*n*=152)
Progressive disease	100 (66%)
Allogeneic HSCT	17 (11%)
Drug related adverse event (Gastrointestinal n=3; cardiac n=1; bleeding n=1; general n=1; haematological n=1; infection n=1; rash n=1)	10 (7%)
Frailty	3 (2%)
Other cancer	2 (1%)
Co-morbidities	1 (<1%)
Death during treatment	18 (12%)

Dose reductions

Of 211 patients nine commenced therapy at an attenuated dose due to frailty or drug interaction. 30 patients (15%) underwent 31 dose reductions during therapy. Reasons for dose reduction were: Haematological toxicity 4, bleeding 4, cardiac 3, fatigue 2, rash 3, cramps 1, arthralgia 1, infection 1, liver dysfunction 2, gastrointestinal toxicity 3, drug interaction 1, frailty 3, unknown 2.

Conclusion

- Despite representing an unselected patient group response rates in our study are equivalent to those described in the pooled analysis[1,2]. Median PFS is modestly reduced (17.8 months vs. 25.4 months), but median OS shows marked divergence (23.9 months vs. 61.6 months). This discrepancy appears most attributable to marked differences in patient demographics (sMIPI high risk 46% in this study vs. 26% in pooled analysis).
- It is reassuring that ibrutinib discontinuation due to toxicity was similar to clinical trial data and incidence of bleeding events leading to alteration in dosing was rare.
- Older patients, often underrepresented in clinical trials, displayed encouraging responses to ibrutinib with patients aged 75 and over displaying median PFS of 11 months comparing favourably with historical registry data[3].
- Use of post-ibrutinib therapy was low overall and outcomes were generally poor indicating an ongoing unmet clinical need in this setting, although outcomes with R-BAC were encouraging.

Disclosures: Janssen provided financial support for study but had no involvement in study design, data analysis or poster design.

References: 1. Rule, S. *et al.* Ibrutinib for the treatment of relapsed/refractory mantle cell lymphoma: extended 3.5-year follow up from a pooled analysis. *Haematologica* 2019; 104:e211-e214. **2.** Rule, S. *et al.* Outcomes in 370 patients with mantle cell lymphoma treated with ibrutinib: a pooled analysis from three open-label studies. Br J Haematol. 2017; 179:430-438. **3.** Smith, A. *et al.* Impact of novel therapies for mantle cell lymphoma in the real world setting: a report from the UK's Haematological Malignancy Research Network (HMRN). *Br J Haematol* 2018;181:215-225.

BSH2020 PO-092-A

Lymphoma/CLL

Rory Mcculloch

Poster presented at: British Society for Haematology Listening • Learning • Leading

PosterSessionOnline.com