Effects of luspatercept on iron overload and impact on responders to luspatercept: results from the BELIEVE trial

John B. Porter,[1] Farrukh Shah,[2] Emma Drasar,[2] Quentin A. Hill,[3] Vip Viprakasit,[4] Ali T. Taher,[5] Pencho Georgiev,[6,7] Kevin H. M. Kuo,[8] Thomas Coates,[9,10] Ersi Voskaridou,[11] Hong Keng Liew,[12] Idit Pazgal-Kobrowski,[13] Gian Luca Forni,[14] Silverio Perrotta,[15] Abderrahim Khelif,[16] Ashutosh Lal,[17] Antonis Kattamis,[18] Abderrahmane Laadem,[19] Jeevan K. Shetty,[20] Peter G. Linde,[21] Olivier Hermine,[22,23] Antonio Piga,[24] Maria Domenica Cappellini[25]

[1]University College London, University College London Hospitals, London, UK; [2]Department of Haematology, Whittington Health NHS Trust, London, UK; [3]St. James University Hospital, Leeds, UK; [4]Siriraj Hospital, Mahidol University, Bangkok, Thailand; [5]Department of Internal Medicine, American University of Beirut Medical Center, Beirut, Lebanon; [6]St George University Hospital for Active Treatment, Plovdiv, Bulgaria; [7]Medical University of Plovdiv, Plovdiv, Bulgaria; [8]Division of Medical Oncology and Hematology, Department of Medicine, University Health Network, and the Division of Hematology, Department of Medicine, University of Toronto, Toronto, Canada; [9]Children's Center for Cancer and Blood Diseases, Children's Hospital Los Angeles, CA, USA; [10]USC Keck School of Medicine, Los Angeles, CA, USA; [11]Thalassemia and Sickle Cell Center of Laiko General Hospital, Athens, Greece; [12]Hospital Sultanah Bahiyah, Alor Setar, Malaysia; [13]Comprehensive Center of Thalassemia, Rabin Medical Center, Beilinson Hospital, Petah Tikva, Israel; [14]Centro della Microcitemia e Anemie Congenite e del Dismetabolismo del Ferro, Ospedale Galliera, Genoa, Italy; [15]Università della Campania, Luigi Vanvitelli, Caserta, Italy; [16]Farhat Hached University Hospital, Sousse, Tunisia; [17]University of California San Francisco Benioff Children's Hospital, Oakland, CA, USA; [18]First Department of Pediatrics, National and Kapodistrian University of Athens, Athens, Greece; [19]Formerly Bristol Myers Squibb, Princeton, NJ, USA; [20]Celgene International Sarl, a Bristol-Myers Squibb Company, Boudry, Switzerland; [21]Formerly Acceleron Pharma, Cambridge, MA, USA; [22]Imagine Institute, Paris, France; [23]Department of Hematology, Necker Hospital, Assistance Publique-Hôpitaux de Paris, Paris, France; [24]Department of Clinical and Biological Sciences, University of Turin, Turin, Italy; [25]Fondazione IRCCS Ca' Granda Policlinico Hospital, University of Milan, Milan, Italy

Introduction

- Patients with β-thalassaemia experience chronic anaemia due to ineffective erythropoiesis as a result of mutations in β-globin[1,2]
- Treatment for anaemia is mainly supportive, in the form of red blood cell (RBC) transfusions; there is a need for new and effective treatments[3]
- Regular RBC transfusions to manage anaemia can lead to iron overload, necessitating iron chelation therapy (ICT)
 - Elevated serum ferritin levels (> 1,000 µg/L) have been associated with significantly increased risk of cardiac failure and death[4]
- Luspatercept is a first-in-class erythroid maturation agent that binds select TGF-β superfamily ligands to diminish Smad2/3 signalling and enhance late-stage erythropoiesis[5,6]
- The phase 3 BELIEVE trial is evaluating the efficacy and safety of luspatercept in adults with β-thalassaemia requiring regular RBC transfusions (NCT02604433)[7]
- In BELIEVE, 21.4% of patients receiving luspatercept achieved the primary endpoint (≥ 33% RBC transfusion burden reduction from baseline during Weeks 13–24), versus 4.5% receiving placebo

Objective

- To evaluate the effects of luspatercept on iron loading over time and the impact of baseline iron parameters on response to luspatercept

Methods

Study design

- The BELIEVE study is an ongoing phase 3, double-blind, randomised, placebo-controlled, multicentre trial
- Adult patients with β-thalassaemia or haemoglobin E/β-thalassaemia (compound β-thalassaemia mutation and/or multiplication of α-globin genes was allowed) requiring regular transfusions of 6–20 RBC units in the 24 weeks prior to randomisation with no transfusion-free period > 35 days were eligible for this study
- Patients were enrolled between July 2016 and June 2017 at 65 sites in 15 countries
- The study was approved by the ethics committee and informed consent was obtained from all participants
- Patients were randomised 2:1 to receive luspatercept (starting dose 1.0 mg/kg, titration allowed to 1.25 mg/kg) or placebo subcutaneously every 21 days for ≥ 48 weeks (**Figure 1**)
- The primary endpoint was achievement of RBC transfusion burden reduction ≥ 33%, with a reduction of ≥ 2 units in Weeks 13–24 versus the 12 weeks prior to randomisation
- After study unblinding, patients randomised to receive placebo were eligible to cross over and be treated with luspatercept

Iron parameters

- Patients were evaluated for changes in the following iron parameters from baseline, with an observation period for up to 96 weeks post-first-dose of luspatercept
 - Serum ferritin levels
 - Liver iron concentration (LIC)
 - Myocardial iron (determined by T2-weighted magnetic resonance imaging [T2* MRI])
- Response to luspatercept by baseline iron parameter subgroup was also assessed

Results

Patients

- Of the 336 patients enrolled, 224 and 112 patients were randomised to receive luspatercept and placebo, respectively (**Table 1**)
- Of the patients in the safety population, 209 of 223 (93.7%) receiving luspatercept and 104 of 109 (95.4%) receiving placebo had a history of iron overload (defined as serum ferritin > 1,000 µg/L or LIC > 7 mg/g dry weight [dw])
- 139 (62.3%), 92 (41.3%), and 83 (37.2%) of 223 patients receiving luspatercept had received prior deferasirox, deferiprone, and deferoxamine, respectively, versus 63 (57.8%), 40 (36.7%), and 39 (35.8%) of 109 in the placebo arm, respectively (**Table 1**)
 - More patients received ICT as monotherapy versus combination therapy (luspatercept arm: 68.8% vs 28.6%; placebo arm: 72.3% vs 24.1%, respectively)
- As of January 7, 2019, a total of 157 (70.4%) of 223 patients initially randomised to receive luspatercept continue to receive active drug and 92 (84.4%) of 109 had crossed over from the placebo arm

Figure 1. Study design of the BELIEVE trial

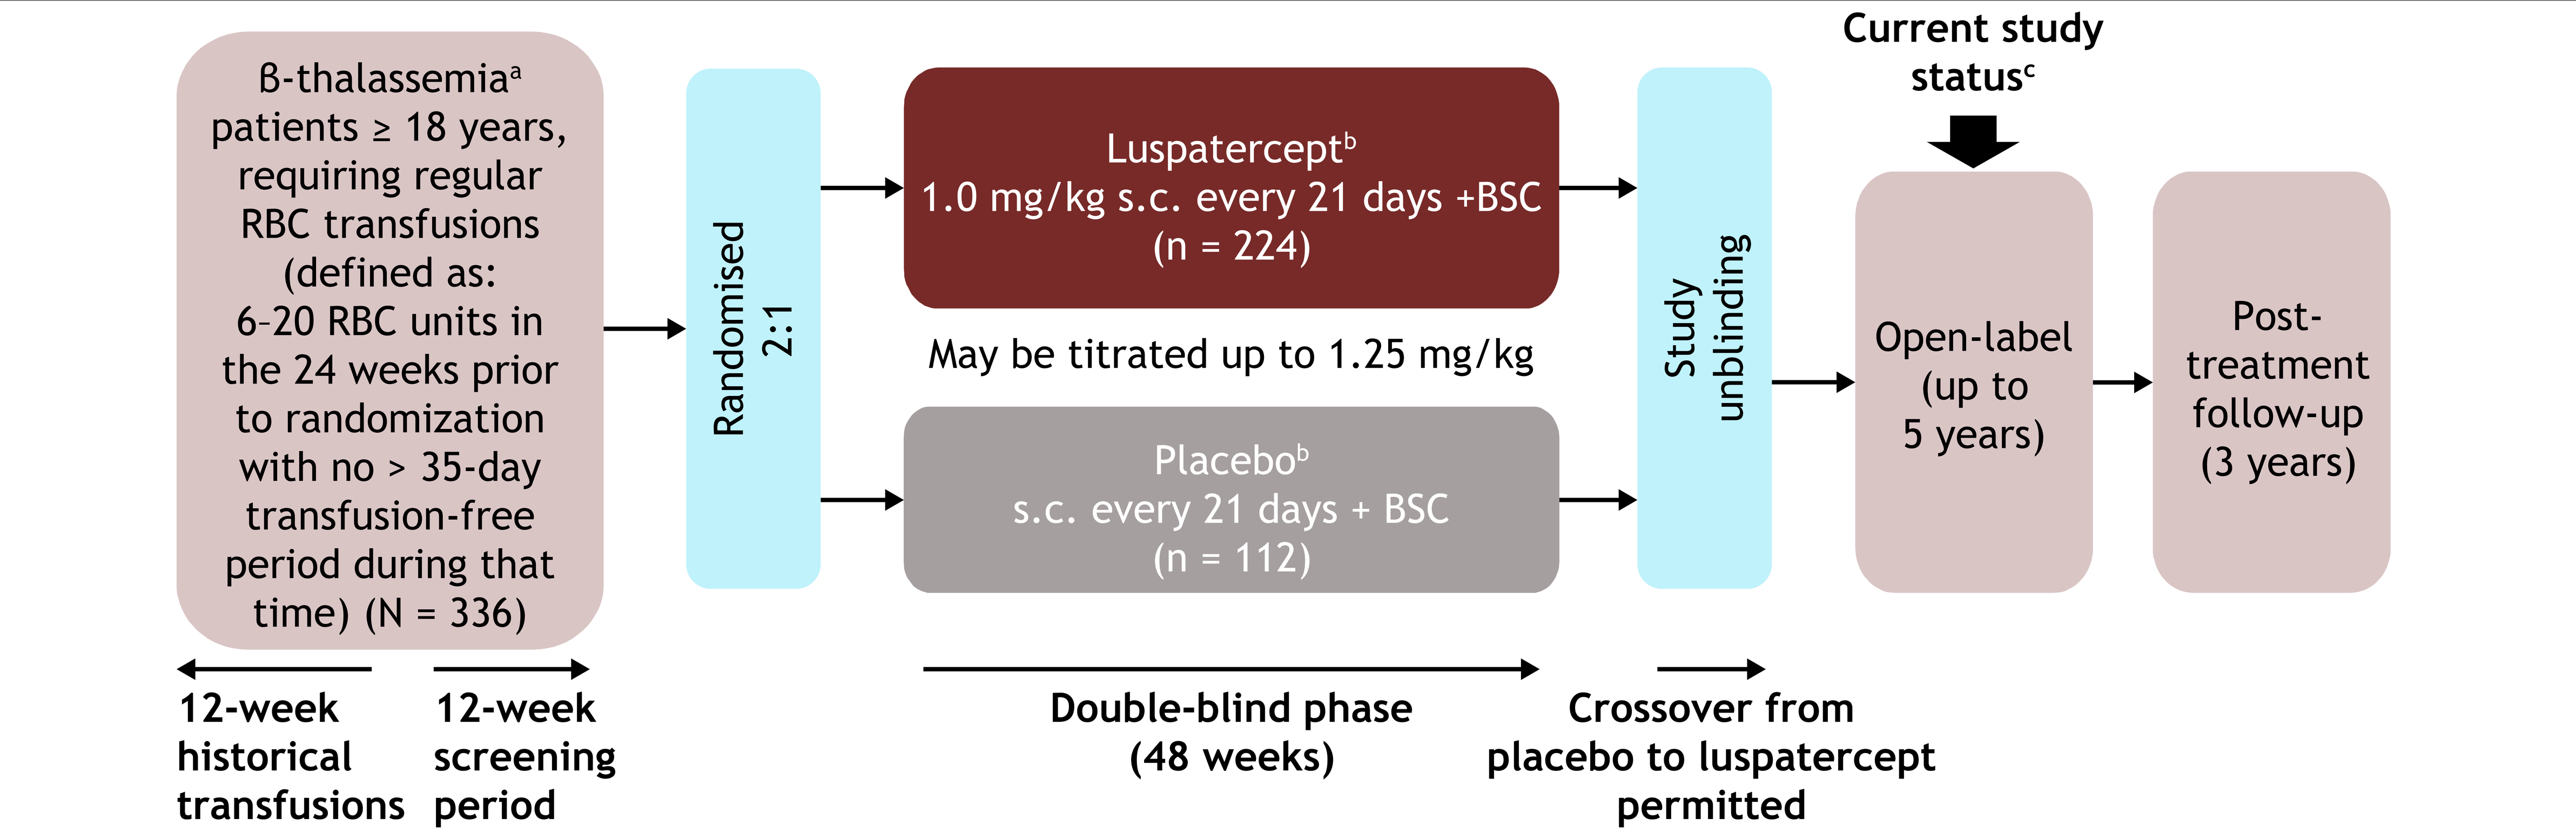
β-thalassemia[a] patients ≥ 18 years, requiring regular RBC transfusions (defined as: 6–20 RBC units in the 24 weeks prior to randomization with no > 35-day transfusion-free period during that time) (N = 336)
Randomised 2:1
Luspatercept[b] 1.0 mg/kg s.c. every 21 days +BSC (n = 224)
May be titrated up to 1.25 mg/kg
Placebo[b] s.c. every 21 days + BSC (n = 112)
Study unblinding
Current study status[c]
Open-label (up to 5 years)
Post-treatment follow-up (3 years)
12-week historical transfusions
12-week screening period
Double-blind phase (48 weeks)
Crossover from placebo to luspatercept permitted

[a]β-thalassaemia or Hb E/β-thalassaemia (compound β-thalassaemia with mutation and/or multiplication of α-globin genes was allowed). [b]Patients could receive RBC transfusions and ICT to maintain their baseline Hb level. [c]The trial is fully enrolled and patients continue to receive treatment or follow-up.
BSC, best supportive care; Hb, haemoglobin; ICT, iron chelation therapy; RBC, red blood cell; s.c., subcutaneously.

Table 1. Baseline characteristics of the BELIEVE ITT population

Characteristic[a]	Luspatercept (N = 224)	Placebo (N = 112)
Age, median (range), years	30 (18-66)	30 (18-59)
Female, n (%)	132 (58.9)	63 (56.3)
$β^0/β^0$, n (%)	68 (30.4)	35 (31.3)
Hb (24 week),[b] median (range), g/dL	9.31 (4.5-11.4)	9.15 (5.8-11.7)
RBC transfusion burden, median (range), units/12 weeks	6.1 (3-14)	6.3 (3-12)
RBC transfusion burden, median (range), units/24 weeks[c]	14 (6-24)	15 (6-26)
Splenectomy, n (%)	129 (57.6)	65 (58.0)
Serum ferritin, mean (SD), µg/L	2,097 (1,757)	1,845 (1,669)
LIC, mean (SD), mg/g dw	12.0 (14.8)	10.1 (11.5)
> 7 mg/g dw, n (%)	103 (46.0)	45 (40.2)
Myocardial iron by T2* MRI, mean (SD), ms	33.5 (16.2)	34.8 (10.7)
Prior iron chelation therapy[c]		
Deferasirox, n (%)	139 (62.3)	63 (57.8)
Deferiprone, n (%)	92 (41.3)	40 (36.7)
Deferoxamine mesylate/deferoxamine, n (%)	83 (37.2)	39 (35.8)

[a]Data on endocrine function were not collected; [b]Defined as the mean of all documented pre-transfusion Hb values during the 24 weeks prior to first dose for each patient. [c]Safety population (luspatercept n = 223; placebo n = 109). dw, dry weight; Hb, hemoglobin; ITT, intent-to-treat; LIC, liver iron concentration; RBC, red blood cell; SD, standard deviation; T2* MRI, T2-weighted magnetic resonance imaging.

Effect of baseline iron parameters on achievement of transfusion burden reduction

- 158 of 224 (70.5%) and 33 of 112 (29.5%) patients in the luspatercept and placebo treatment arms, respectively, achieved a ≥ 33% reduction in RBC transfusion burden with a reduction of ≥ 2 RBC units during any 12-week interval in the first 48 weeks
 - Response by baseline iron parameters at Weeks 48 and 96 is shown in **Table 2**
- Responses to luspatercept were achieved regardless of baseline iron parameter thresholds

Table 2. Achievement of ≥ 33% reduction in RBC transfusion burden over any 12 weeks by baseline serum ferritin level, LIC, and myocardial iron

Characteristic, n/N (%)	Assessed over 48 weeks[a]		Assessed over 96 weeks[b]	
	Luspatercept (N = 224)	Placebo (N = 112)	Luspatercept (N = 224)	Placebo (N = 112)
Baseline serum ferritin level, µg/L	**n = 207**	**n = 101**	**n = 165**	**n = 101**
< 1,000	54/73 (74.0)	11/45 (24.4)	53/67 (79.1)	14/45 (31.1)
≥ 1,000	97/134 (72.4)	21/56 (37.5)	77/98 (78.6)	24/56 (42.9)
Baseline LIC, mg/g dw	**n = 202**	**n = 103**	**n = 88**	**n = 3**
≤ 3	52/67 (77.6)	9/35 (25.7)	22/24 (91.7)	0/3
> 3 to ≤ 7	37/47 (78.7)	11/29 (37.9)	18/21 (85.7)	0/0
> 7 to ≤ 15	31/36 (86.1)	11/17 (64.7)	15/18 (83.3)	0/0
> 15	39/52 (75.0)	7/22 (31.8)	21/25 (84.0)	0/0
Myocardial iron T2* MRI, ms	**n = 201**	**n = 102**	**n = 91**	**n = 3**
≤ 10	6/8 (75.0)	2/3 (66.7)	4/4 (100)	0/0
> 10 to ≤ 20	17/22 (77.3)	1/8 (12.5)	9/10 (90.0)	0/0
> 20	136/171 (79.5)	34/91 (37.4)	67/77 (87.0)	0/3

[a]During the 48-week double-blind treatment period. [b]Includes the 48-week double-blind treatment period and beyond.
dw, dry weight; LIC, liver iron concentration; RBC, red blood cell; T2* MRI, T2-weighted magnetic resonance imaging.

Change in iron parameters from baseline to Week 96

- Change from baseline in iron parameters was evaluated over the last 24 weeks prior to Week 96 (**Table 3**) (**Figure 2**)
- Change in iron parameters from baseline was also evaluated in patients achieving ≥ 33% reduction in transfusion burden over any 12 weeks with available Week 96 data (**Figure 2**):
 - Among the 130 (78.8%) luspatercept- and 38 (37.6%) placebo-treated responders over any 12 weeks, mean (standard deviation [SD]) serum ferritin change from baseline was −490 (830) µg/L and +162 (780) µg/L, respectively (least square mean difference: −665.94; *P* = 0.0001)
 - In luspatercept non-responders, a decrease in serum ferritin was still observed, although the magnitude of decrease (−122.29 µg/L) was smaller compared with the responders
 - In luspatercept responders, mean LIC decreases were more pronounced than in all patients; mean (SD) changes were −1.33 (8.22) mg/g dw and −1.82 (9.03) mg/g dw among patients achieving ≥ 33% transfusion burden reduction for ≥ 12 and ≥ 24 weeks, respectively
 - Although the greatest changes were observed in responding patients, overall there were clinically meaningful reductions in patients receiving luspatercept
 - Among the 80 (87.9%) luspatercept responders, mean changes from baseline in myocardial iron T2* were within the normal myocardial iron range and had little clinical significance
- There was no significant decrease from baseline in ICT mean daily dose between luspatercept- and placebo-treated patients
 - In responding patients (those who achieved ≥ 33% reduction in transfusion burden/24 weeks) there was a more pronounced decrease in deferiprone mean daily dose in luspatercept (−831.9 mg) versus all luspatercept- and placebo-treated patients (−325.3 and −301.7 mg, respectively)

Table 3. Mean change in iron parameters for all patients at Week 96

Characteristic	Luspatercept	Placebo	LSM difference	*P* value
Serum ferritin level	**n = 165**	**n = 101**		
Change from baseline, mean (SD), µg/L	−412 (892)	+95 (656)	−503.23	**< 0.001**
LIC	n = 88	n = 3		
Change from baseline, mean (SD), mg/g dw	**−0.38 (8.40)**	**+1.86 (2.07)**	**−1.83**	**0.6901**
Myocardial iron T2* MRI	n = 91	n = 3		
Change from baseline, mean (SD), ms	−0.39 (19.73)	−2.38 (5.15)	−2.95	0.6101

dw, dry weight; LIC, liver iron concentration; LSM, least square mean; SD, standard deviation; T2* MRI, T2-weighted magnetic resonance imaging.

Figure 2. Change in iron parameters from baseline[a,b]

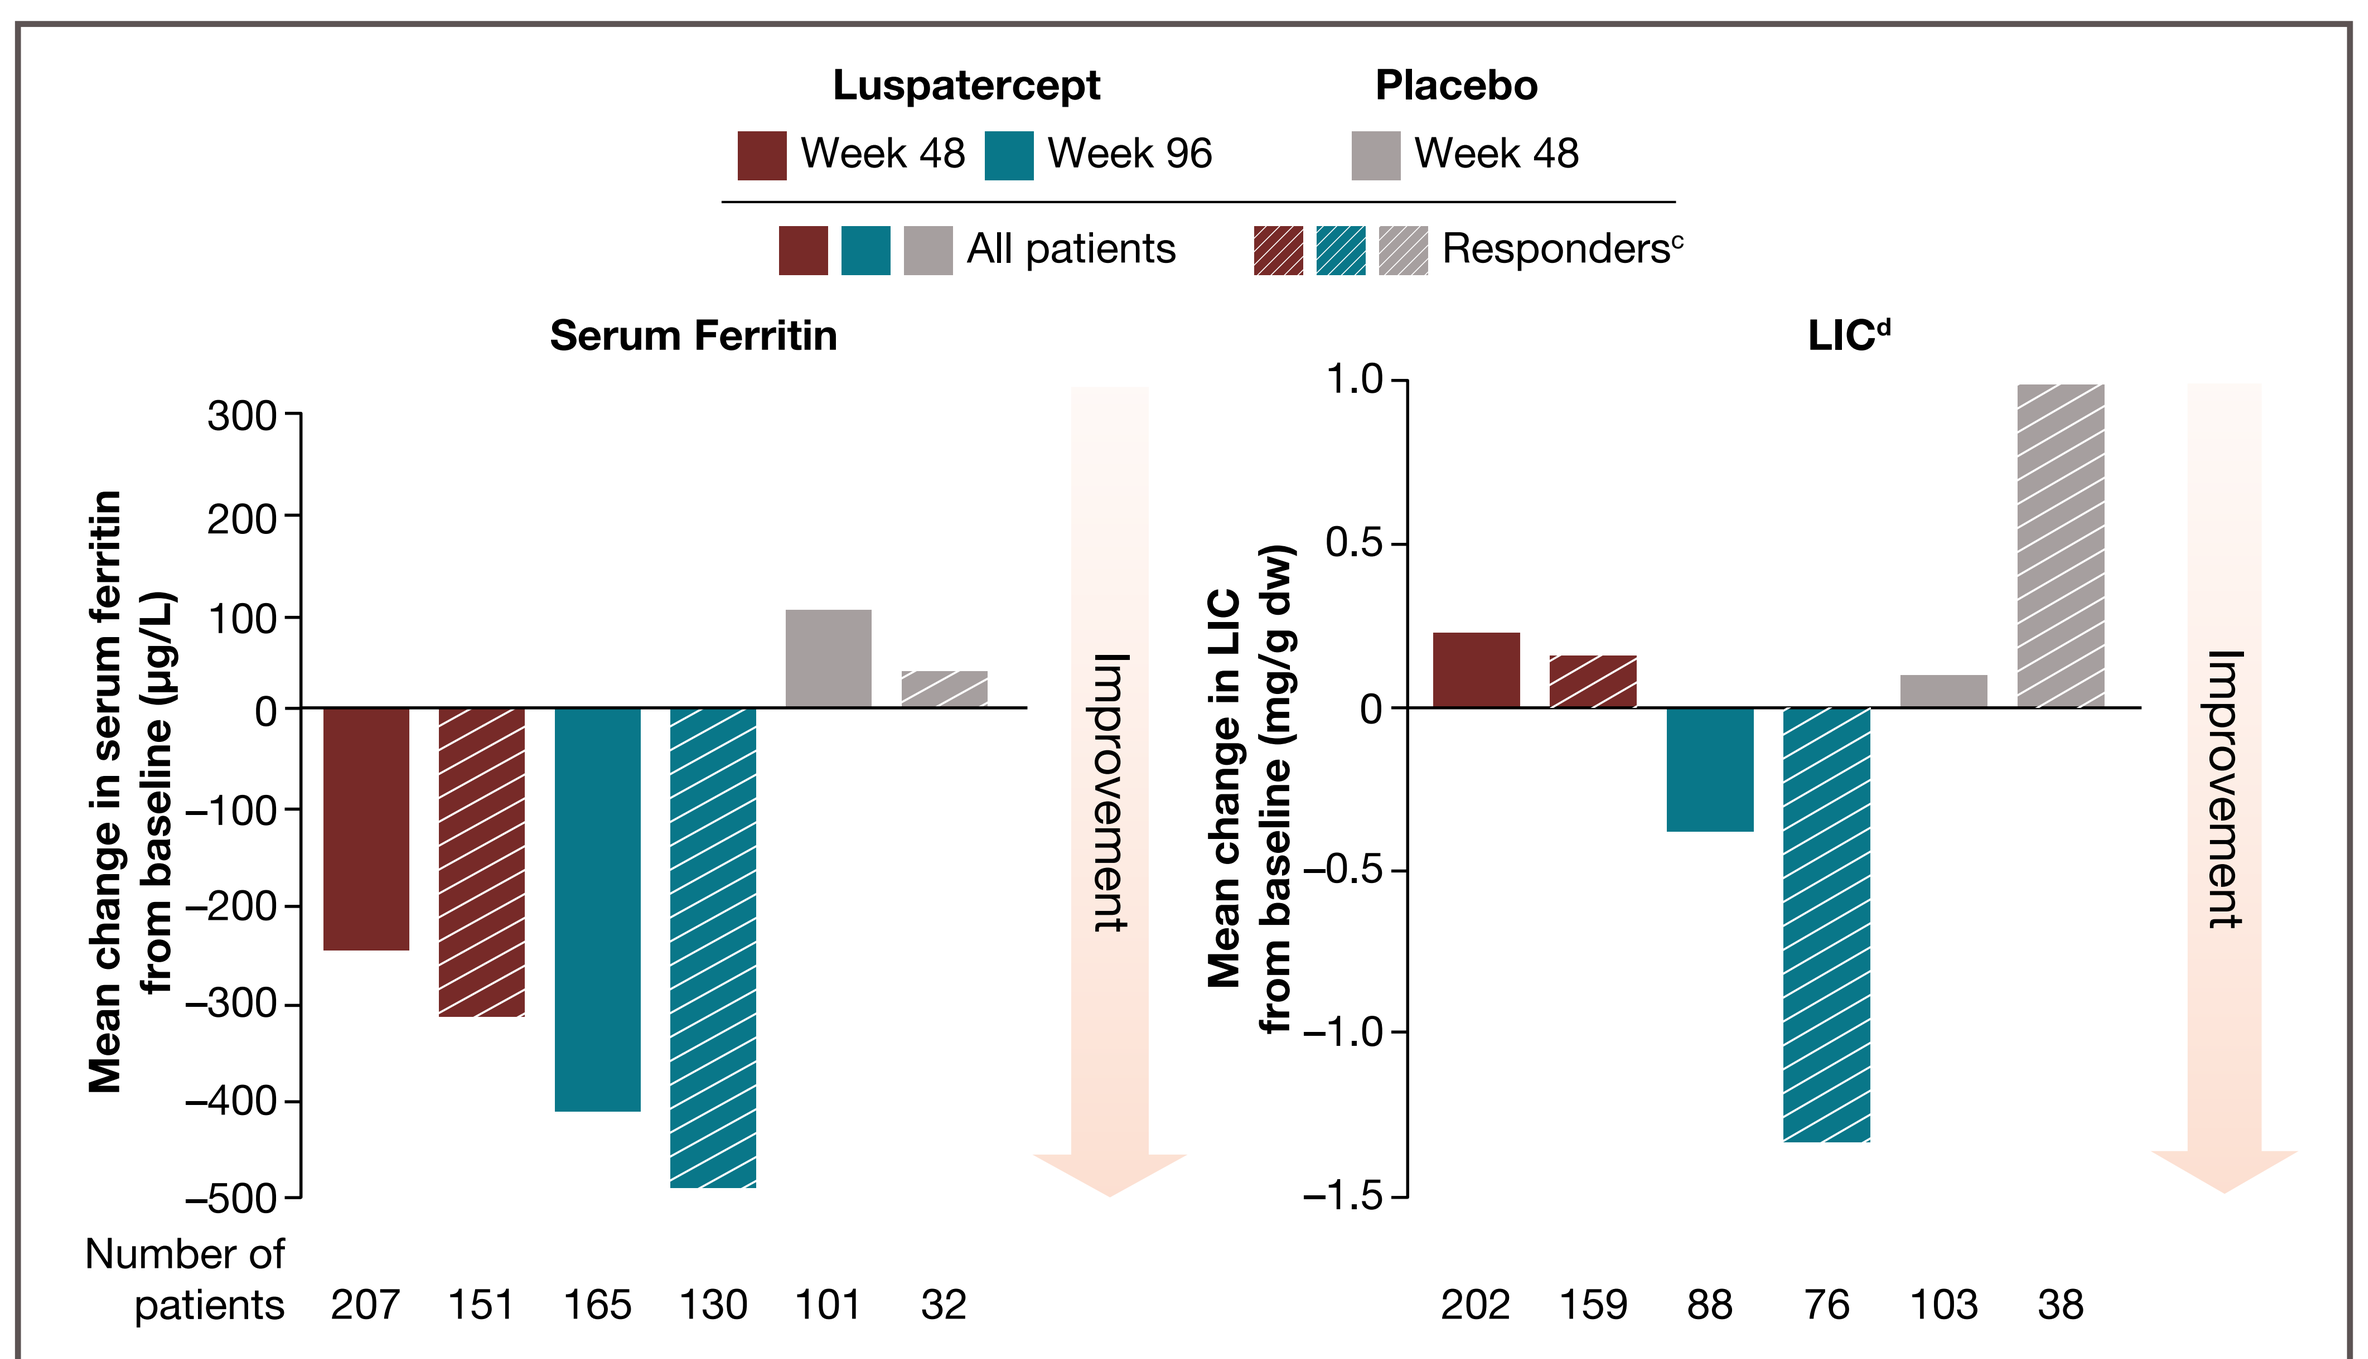
Luspatercept
Week 48
Week 96
Placebo
Week 48
All patients
Responders[c]
Serum Ferritin
Mean change in serum ferritin from baseline (µg/L)
300
200
100
0
−100
−200
−300
−400
−500
Improvement
Number of patients
207
151
165
130
101
32
LIC[d]
Mean change in LIC from baseline (mg/g dw)
1.0
0.5
0
−0.5
−1.0
−1.5
Improvement
202
159
88
76
103
38

[a]Week 48 and Week 96 measurements were the mean of measurements made within the last 24 weeks of the 48- or 98-week treatment period, respectively. [b]Mean reductions in myocardial iron with luspatercept remained within the normal clinical range and were not considered clinically meaningful (baseline mean myocardial iron T2* MRI values were 33.52 ms and 34.76 ms for luspatercept- and placebo-treated patients, respectively). [c]Defined as achievement of ≥ 33% reduction in transfusion burden over any 12 weeks. [d]Measurements were made locally and analysis was corrected using a standardised method.
dw, dry weight; LIC, liver iron concentration; T2* MRI, T2-weighted magnetic resonance imaging.

Change in patient iron parameter categories from baseline to Week 96

- Of the 98 luspatercept-treated patients with baseline mean serum ferritin ≥ 1,000 µg/L, 42 (42.9%) achieved post-baseline mean serum ferritin of < 1,000 µg/L, when assessed over the 24 weeks prior to Week 96
- 3 (21.4%) luspatercept-treated patients with baseline myocardial iron T2* MRI ≤ 20 ms had post-baseline T2* > 20 ms
- 11 (44%) patients receiving luspatercept with baseline LIC > 15 mg/g dw had post-baseline LIC ≤ 15 mg/g dw

Conclusions

- Luspatercept treatment resulted in clinically meaningful and maintained reductions in serum ferritin levels
- Baseline iron overload did not seem to affect response rates with luspatercept
- Treatment resulted in clinically meaningful reductions in RBC transfusion burden regardless of baseline serum ferritin level
- There was a trend for decrease in LIC with longer follow up at 96 weeks; among responders, the decrease was more pronounced compared with non-responders

References

1. Camaschella C, Nai A. *Br J Haematol* 2016;172:512-523.
2. Taher AT, et al. *Lancet* 2018;391:155-167.
3. Taher AT, et al. *Blood* 2018;132:1781-1791.
4. Borgna-Pignatti C, et al. *Haematologica* 2004;89:1187-1193.
5. Attie KM, et al. *Am J Hematol* 2014;89:766-770.
6. Suragani RN, et al. *Nat Med* 2014;20:408-414.
7. Cappellini MD, et al. *N Engl J Med* 2020;382:1219-1231.

Acknowledgments

- The study was supported by Celgene, a Bristol-Myers Squibb Company, in collaboration with Acceleron Pharma
- All authors contributed to and approved the presentation; writing and editorial assistance were provided by Daniel Gilmartin, PhD, of Excerpta Medica, funded by Bristol Myers Squibb

Disclosures

J. Porter Conflict with: Agios, Bluebird Bio, Celgene (now BMS) (consultancy, honoraria), Conflict with: La Jolla, Protagonist, Silence Therapeutics, Vifor (honoraria), F. Shah Conflict with: Abfero Pharmaceuticals (clinical trial safety committee); Celgene (now BMS) (membership on an entity's steering committee); Silence Therapeutics, Roche, Novartis (membership on an entity's board of directors or advisory committees); Bluebird Bio, E. Drasar Conflict with: Novartis (advisory panel, speaking commitments), Q. A. Hill Conflict with: Novartis, V. Viprakasit Conflict with: Agios, Ionis, La Jolla, Protagonist (consultancy, research funding), Conflict with: Celgene (now BMS) (consultancy, honoraria, research funding); Novartis (consultancy, honoraria, research funding, speakers bureau), A. Taher Conflict with: Celgene (now BMS), La Jolla Pharmaceuticals, Novartis (consultancy, research funding); Ionis Pharmaceuticals, Vifor (consultancy), Conflict with: Protagonist Therapeutics (research funding), Conflict with: Novartis (honoraria), P. Georgiev: None Declared, K. H. M. Kuo Conflict with: Agios, Apellis, Bluebird Bio, Celgene (now BMS), Pfizer (consultancy); Alexion, Novartis (consultancy, honoraria), Conflict with: Bioverativ (data safety monitoring board), T. Coates Conflict with: Apo Pharma (consultancy, honoraria, speakers bureau); Agios Pharma (consultancy, honoraria); Celgene (now BMS) (consultancy, honoraria, steering committee of clinical study), E. Voskaridou Conflict with: Acceleron Pharma, Celgene (now BMS), Genesis (consultancy, research funding), Conflict with: Protagonist (research funding), Conflict with: Addmedica (membership on a board of directors or advisory committee), H. K. Liew: None Declared, I. Pazgal-Kobrowski: None Declared, G. L. Forni Conflict with: Novartis, Celgene (now BMS), Bluebird Bio, Roche (consultancy), Conflict with: Apothex (data and safety monitoring board), S. Perrotta Conflict with: Novartis (research funding, honoraria); Acceleron Pharma (research funding), A. Khelif: None Declared, A. Lal Conflict with: Bluebird Bio, Insight Magnetics, La Jolla Pharmaceutical Company, Novartis, Prognostic Therapeutics, Terumo BCT (grant/contract); Celgene (now BMS) (grant/contract, consultant), A. Kattamis Conflict with: Novartis (honoraria, membership on an entity's board of directors or advisory committee, research funding); Celgene (now BMS) (honoraria, membership on an entity's board of directors or advisory committee); Apopharma (honoraria); Vertex, Vifor, Ionis (membership on an entity's board of directors or advisory committee), A. Laadem Conflict with: Celgene (now BMS) (employment, equity ownership), J. K. Shetty Conflict with: Celgene (now BMS) (employment, equity ownership), P. G. Linde Conflict with: Acceleron Pharma (employment, equity ownership); Fibrogen Inc., Abbott Laboratories (equity ownership), O. Hermine Conflict with: AB Science (consultancy, equity ownership, honoraria, research funding), Conflict with: Celgene (now BMS), Novartis (research funding), A. Piga Conflict with: Celgene (now BMS), Novartis (consultancy, research funding), Conflict with: Acceleron Pharma (research funding), M. D. Cappellini Conflict with: Celgene (now BMS) (honoraria); Genzyme/Sanofi (honoraria, membership on a board of directors or advisory committee); Novartis, Vifor, CRISPR Therapeutics (membership on a board of directors or advisory committee)

Presented at the 60th Annual Scientific Meeting of the British Society for Haematology (BSH); 9–14 November 2020.

John Porter – j.porter@ucl.ac.uk

BSH2020
PO-143-B
Red Cell Disorders
Jennifer Mark
Poster presented at: British Society for Haematology
PosterSessionOnline.com