PO-124

INSIGHT MM Study of Routine Clinical Practice in Multiple Myeloma: Analysis of Duration of Therapy and Treatment Sequencing in UK Patients

Gordon Cook,[1] John Ashcroft,[2] Kevin Boyd,[3] Mamta Garg,[4] Rachel Hall,[5] Bhuvan Kishore,[6] Sally Moore,[7] Dean Smith,[8] Nicola Crofts,[9] Kaili Ren,[10] Guy Pratt[6]

[1]Leeds Teaching Hospitals NHS Foundation Trust, Leeds, UK; [2]Mid Yorkshire Hospitals NHS Foundation Trust, Wakefield, UK; [3]The Royal Marsden NHS Foundation Trust, London, UK; [4]Department of Haematology, University Hospitals of Leicester NHS Trust, Leicester, UK; [5]Royal Bournemouth Hospital NHS Foundation Trust, Bournemouth, UK; [6]University Hospitals Birmingham NHS Foundation Trust, Birmingham, UK; [7]Royal United Hospitals Bath NHS Foundation Trust, Bath, UK; [8]Nottingham University Hospitals NHS Trust, Nottingham, UK; [9]Takeda UK Limited, High Wycombe, UK; [10]Millennium Pharmaceuticals, Inc., Cambridge, MA, USA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited.

Background

- Treatment pathways for patients with NDMM and RRMM vary worldwide, with no current global standard of care.
- Randomised controlled clinical trial data inevitably inform treatment approval and access; however, real-world data are becoming increasingly important to fully define treatment effectiveness in the clinical setting.

Continuous therapy and the gap between efficacy and effectiveness

- In a pooled analysis of patients with NDMM from GIMEMA-MM-03-05, RV-MM-PI-209 and CC-5013-MM-015, continuous therapy significantly improved PFS versus fixed duration of therapy.[1]
- Despite clinical trials showing benefits of continuous versus fixed duration therapy in MM,[1] shorter treatment durations are seen in the real world.[2]
- Differences may be due to:
 - Patient selection[2]
 - Treatment centre effect
 - Study design and protocol rigor
 - Physician/patient preference
 - Burden of treatment.[3]
- Furthermore, 40% of patients do not meet standard clinical trial eligibility criteria.[3,4]

INSIGHT MM (NCT02761187)

- Largest global, prospective, non-interventional, observational study of patients with NDMM/RRMM to date.

Primary objective[5]

- Describe contemporary, real-world patterns of patient characteristics, clinical disease presentation, therapeutic regimen chosen and clinical outcomes in patients with NDMM and patients with RRMM.

Secondary objectives[5]

- Describe patient characteristics, clinical disease presentation, therapeutic regimen chosen and clinical outcomes in NDMM and RRMM patients by type of treatment facility and country.
- Describe patterns and durations of treatment combinations, sequencing, retreatment and continuous versus fixed duration treatment strategies, and the clinical outcomes associated with different treatment regimens.
- Describe factors associated with treatment initiation, treatment modification or treatment change over time, including whether treatment at relapse was initiated due to biochemical progression versus symptomatic progression.
- Describe HRQoL and healthcare resource utilisation.
- Explore associations between patient characteristics, clinical disease presentation, therapeutic regimen chosen and clinical outcomes.

Study aims

- Analyse the UK cohort of the INSIGHT MM study.
- Evaluate DOT, reasons for discontinuation and subsequent treatments in patients with NDMM/RRMM treated with selected regimens at first, second or third LOT.

Methods

- The study design, key eligibility criteria and assessments are summarised in **Figure 1**.[5]

Figure 1. Study design[5]

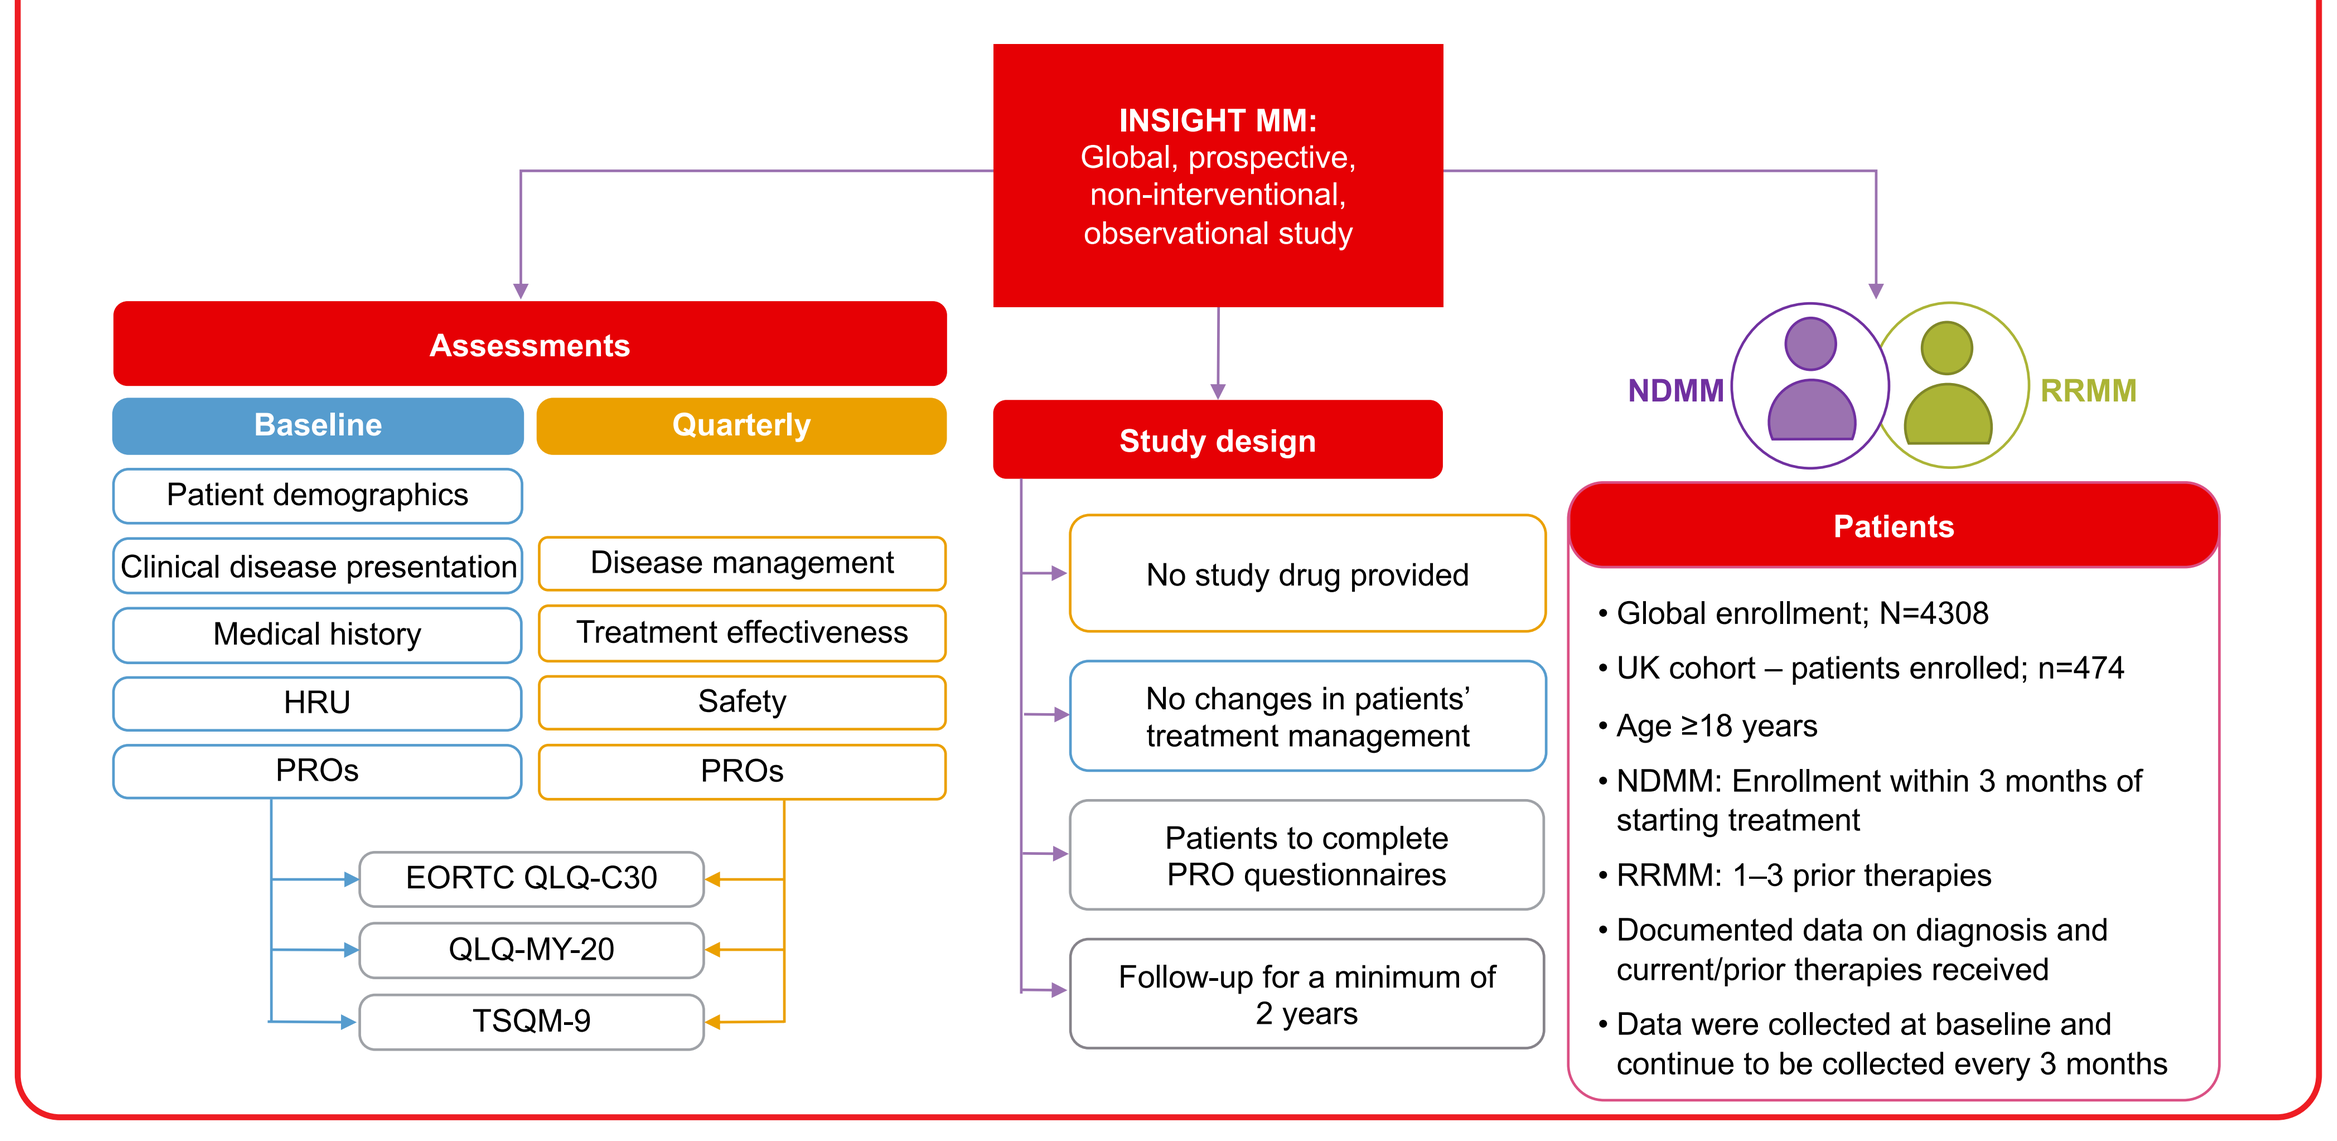
INSIGHT MM:
Global, prospective, non-interventional, observational study
Assessments
Baseline
Quarterly
Patient demographics
Clinical disease presentation
Disease management
Medical history
Treatment effectiveness
HRU
Safety
PROs
PROs
EORTC QLQ-C30
QLQ-MY-20
TSQM-9
Study design
No study drug provided
No changes in patients' treatment management
Patients to complete PRO questionnaires
Follow-up for a minimum of 2 years
NDMM
RRMM
Patients
• Global enrollment; N=4308
• UK cohort – patients enrolled; n=474
• Age ≥18 years
• NDMM: Enrollment within 3 months of starting treatment
• RRMM: 1–3 prior therapies
• Documented data on diagnosis and current/prior therapies received
• Data were collected at baseline and continue to be collected every 3 months

Results

Patient characteristics

- The UK enrolled 474 patients (11% of the INSIGHT MM study).
- This analysis includes 373 of these patients.
- Baseline characteristics of these patients are shown in **Table 1**.
- For this data cut-off (November 2016–November 2018; median follow-up, 12.3 months [range, 0.4–24.4]), 126 patients with NDMM and 247 with RRMM had been enrolled, including 161 and 48 of the total patient population who had reached second- and third-line therapy, respectively.
- The majority of patients were male; median age of overall population was 67 years.
- Similar proportions of patients were ISS disease stage I, II or III at diagnosis.
- More patients with RRMM than NDMM had a prior history of peripheral neuropathy.

Table 1. Patient demographics and baseline characteristics

Characteristic	NDMM* (n=126)	RRMM* (n=247)	Total (N=373)
Male/female, %	53/47	67/33	62/38
Median age, years (range)	67 (38–89)	68 (39–87)	67 (38–89)
Median time from initial diagnosis to study entry, months (range)	2 (0–81)	53 (7–378), [n=241]	38 (0–378), [n=367]
Creatinine clearance (<30 mL/min), %	11	5	7
Elevated calcium (>11 mg/dL), %	8	7	7
Bone lesions present at diagnosis, %	52	33	39
Haemoglobin, n (%)			
Males [females]			
≥12 g/dL [≥11 g/dL]	28 (41.8) [25 (42.4)]	35 (21.3) [31 (37.8)]	63 (27.3) [56 (39.7)]
<12 g/dL [<11 g/dL]	36 (53.7) [31 (52.5)]	66 (40.2) [30 (36.6)]	102 (44.2) [61 (43.3)]
Not available/missing, n	3 (4.5)/0 [3 (5.1)/0]	63 (38.4)/1 [21 (25.6)/0]	66 (28.6)/1 [24 (17.0)/0]
ISS disease stage at initial diagnosis, %			
I	29	12	18
II	25	18	21
III	21†	14‡	16§
History of peripheral neuropathy, %	14	44	34
Type of treatment facility, %			
Academic/university	71	82	78
Community	29	18	22

*Data shown are as at study entry unless otherwise stated,
†25% unavailable/missing data.
‡56% unavailable/missing data.
§45% unavailable/missing data.

Treatment heterogeneity

- Regimen use in patients with NDMM is shown by SCT candidate status in **Figure 2**.
- Treatment regimens most used in patients with RRMM, by LOT are shown in **Figure 3**.

Figure 2. Regimen use in patients with NDMM by SCT candidate status

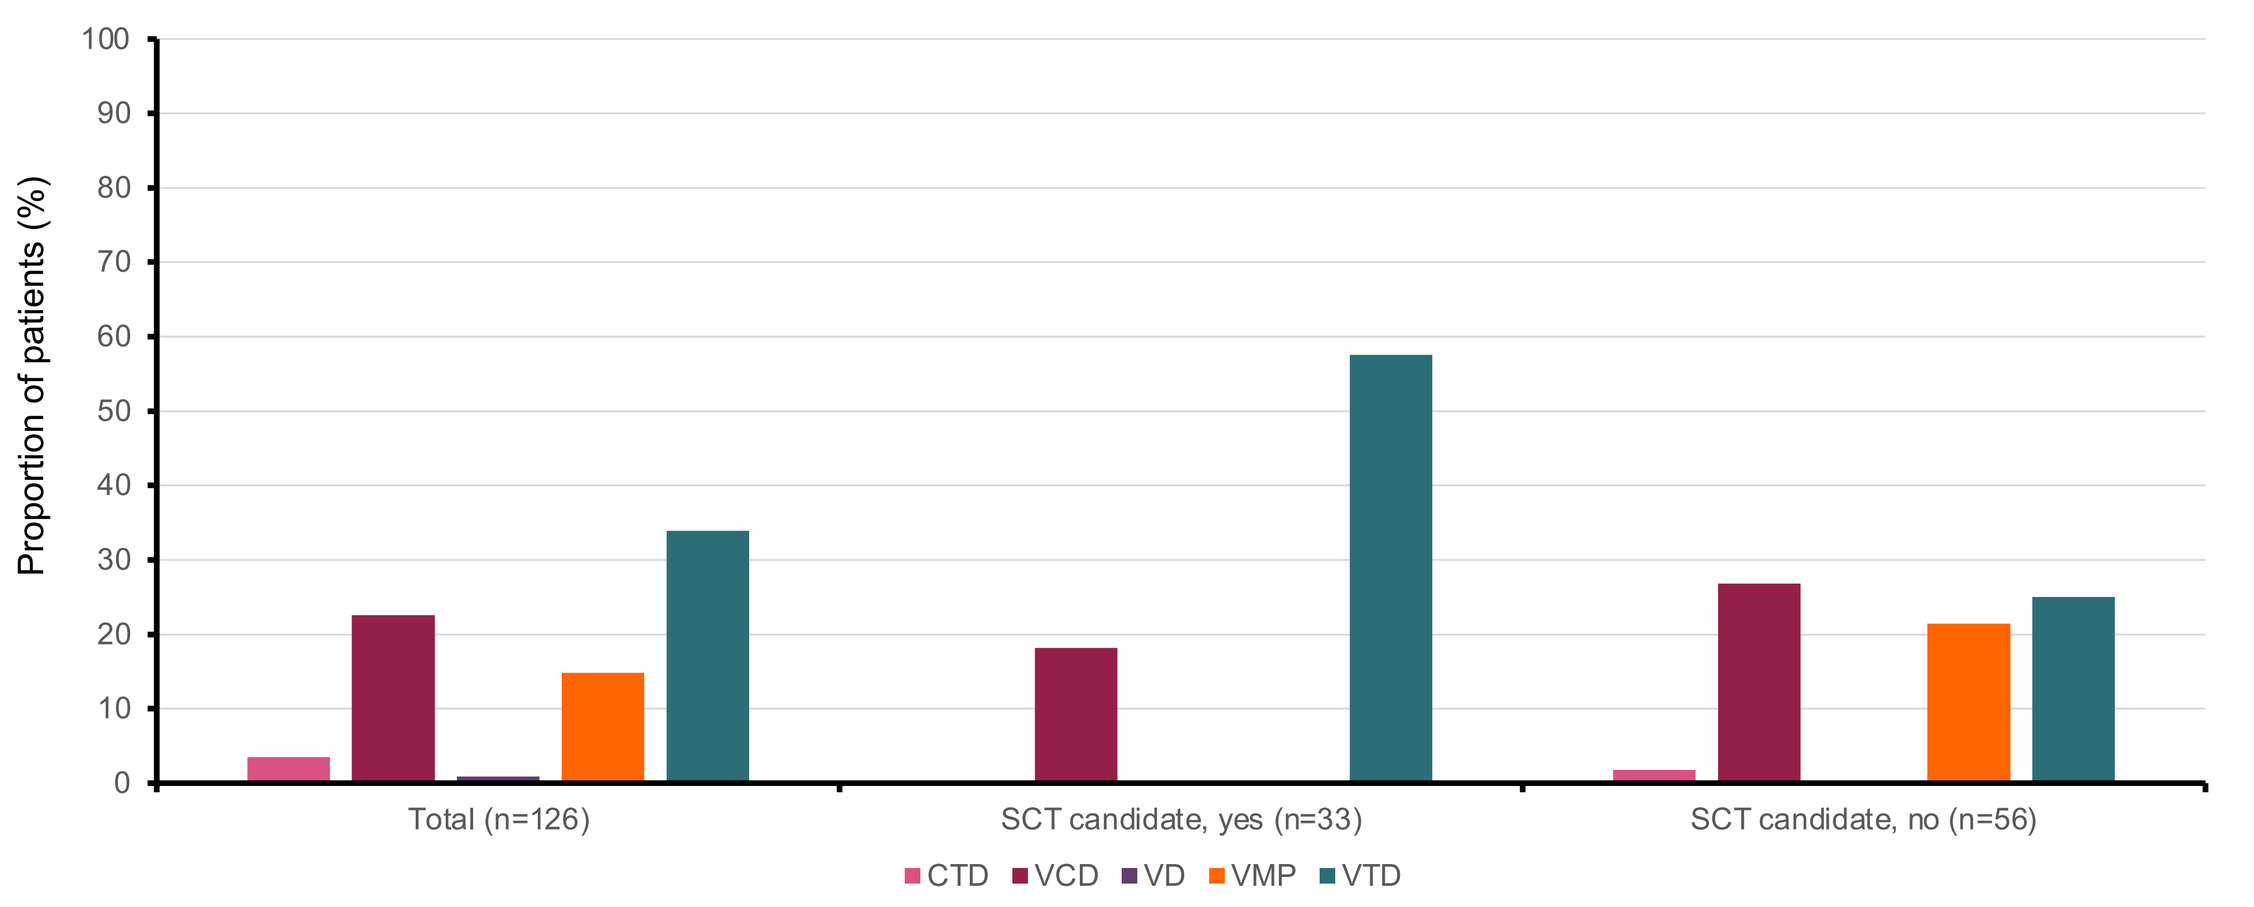
Proportion of patients (%)
Total (n=126)
SCT candidate, yes (n=33)
SCT candidate, no (n=56)
CTD VCD VD VMP VTD

Figure 3. Most common regimens by LOT in patients with RRMM

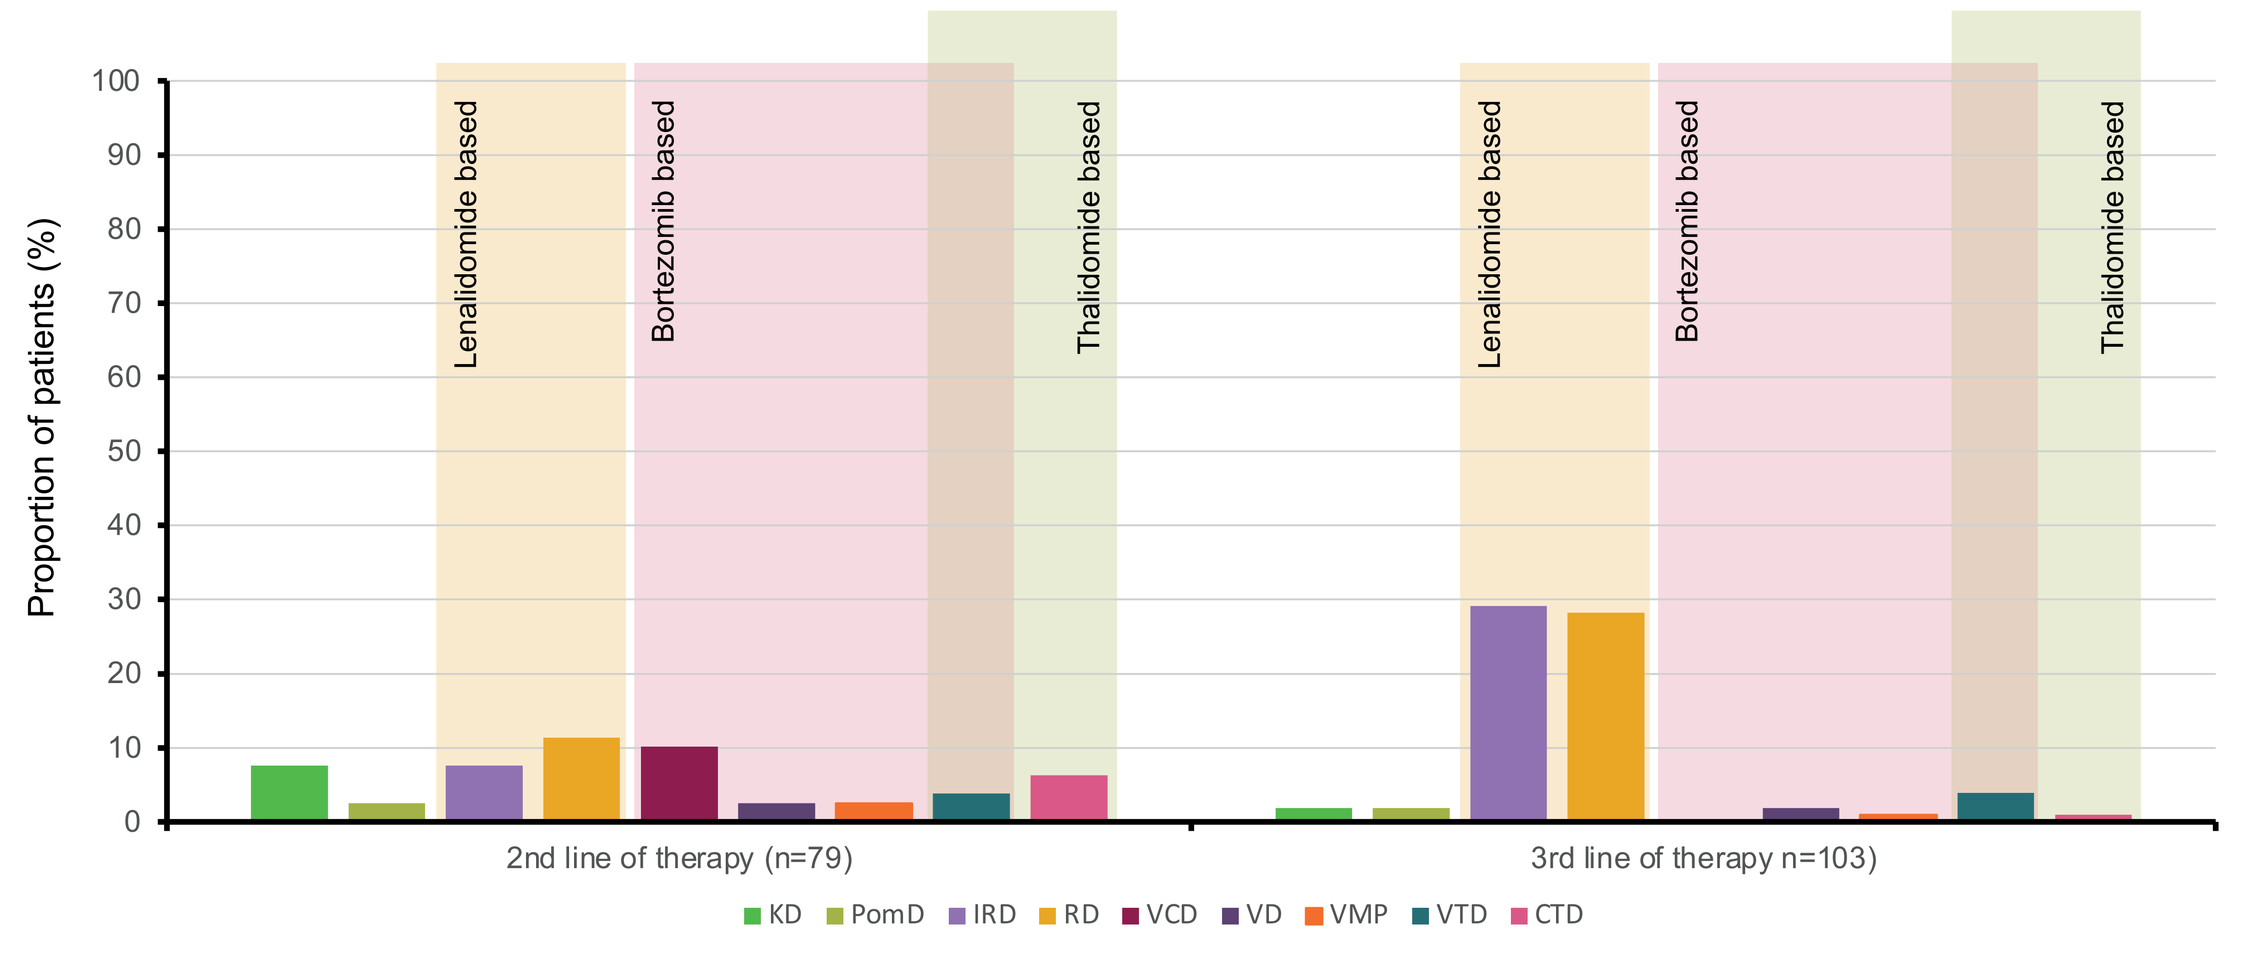
Proportion of patients (%)
Lenalidomide based
Bortezomib based
Thalidomide based
2nd line of therapy (n=79)
3rd line of therapy n=103)
KD PomD IRD RD VCD VD VMP VTD CTD

Treatment discontinuation

- Treatment discontinuation is shown for each regimen and for each LOT in **Figure 4**.
- Median time to regimen discontinuation and duration of treatment for each regimen are shown in **Table 2**.

Figure 4a. Completed, discontinued or ongoing therapies – by regimen (any LOT)

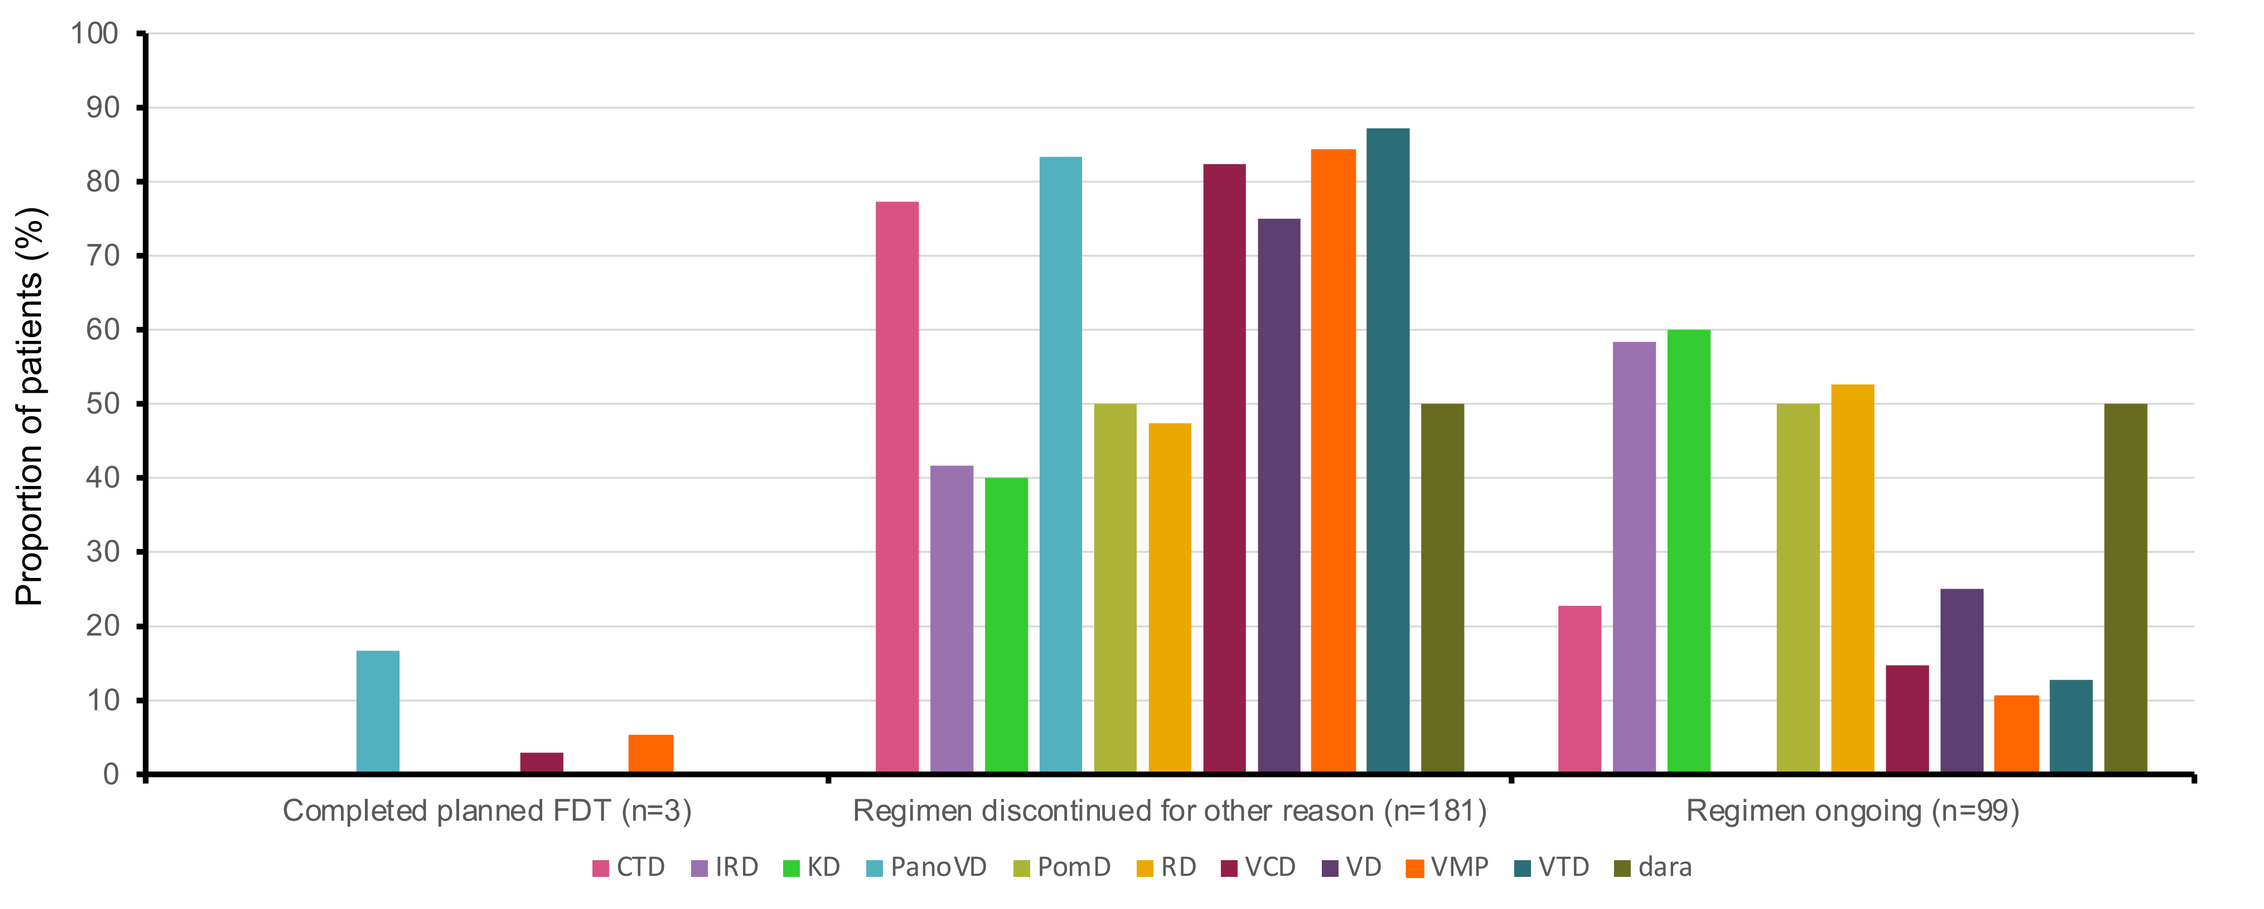
Proportion of patients (%)
Completed planned FDT (n=3)
Regimen discontinued for other reason (n=181)
Regimen ongoing (n=99)
CTD IRD KD PanoVD PomD RD VCD VD VMP VTD dara

Figure 4b. Completed, discontinued or ongoing therapies – by regimen (LOT1)

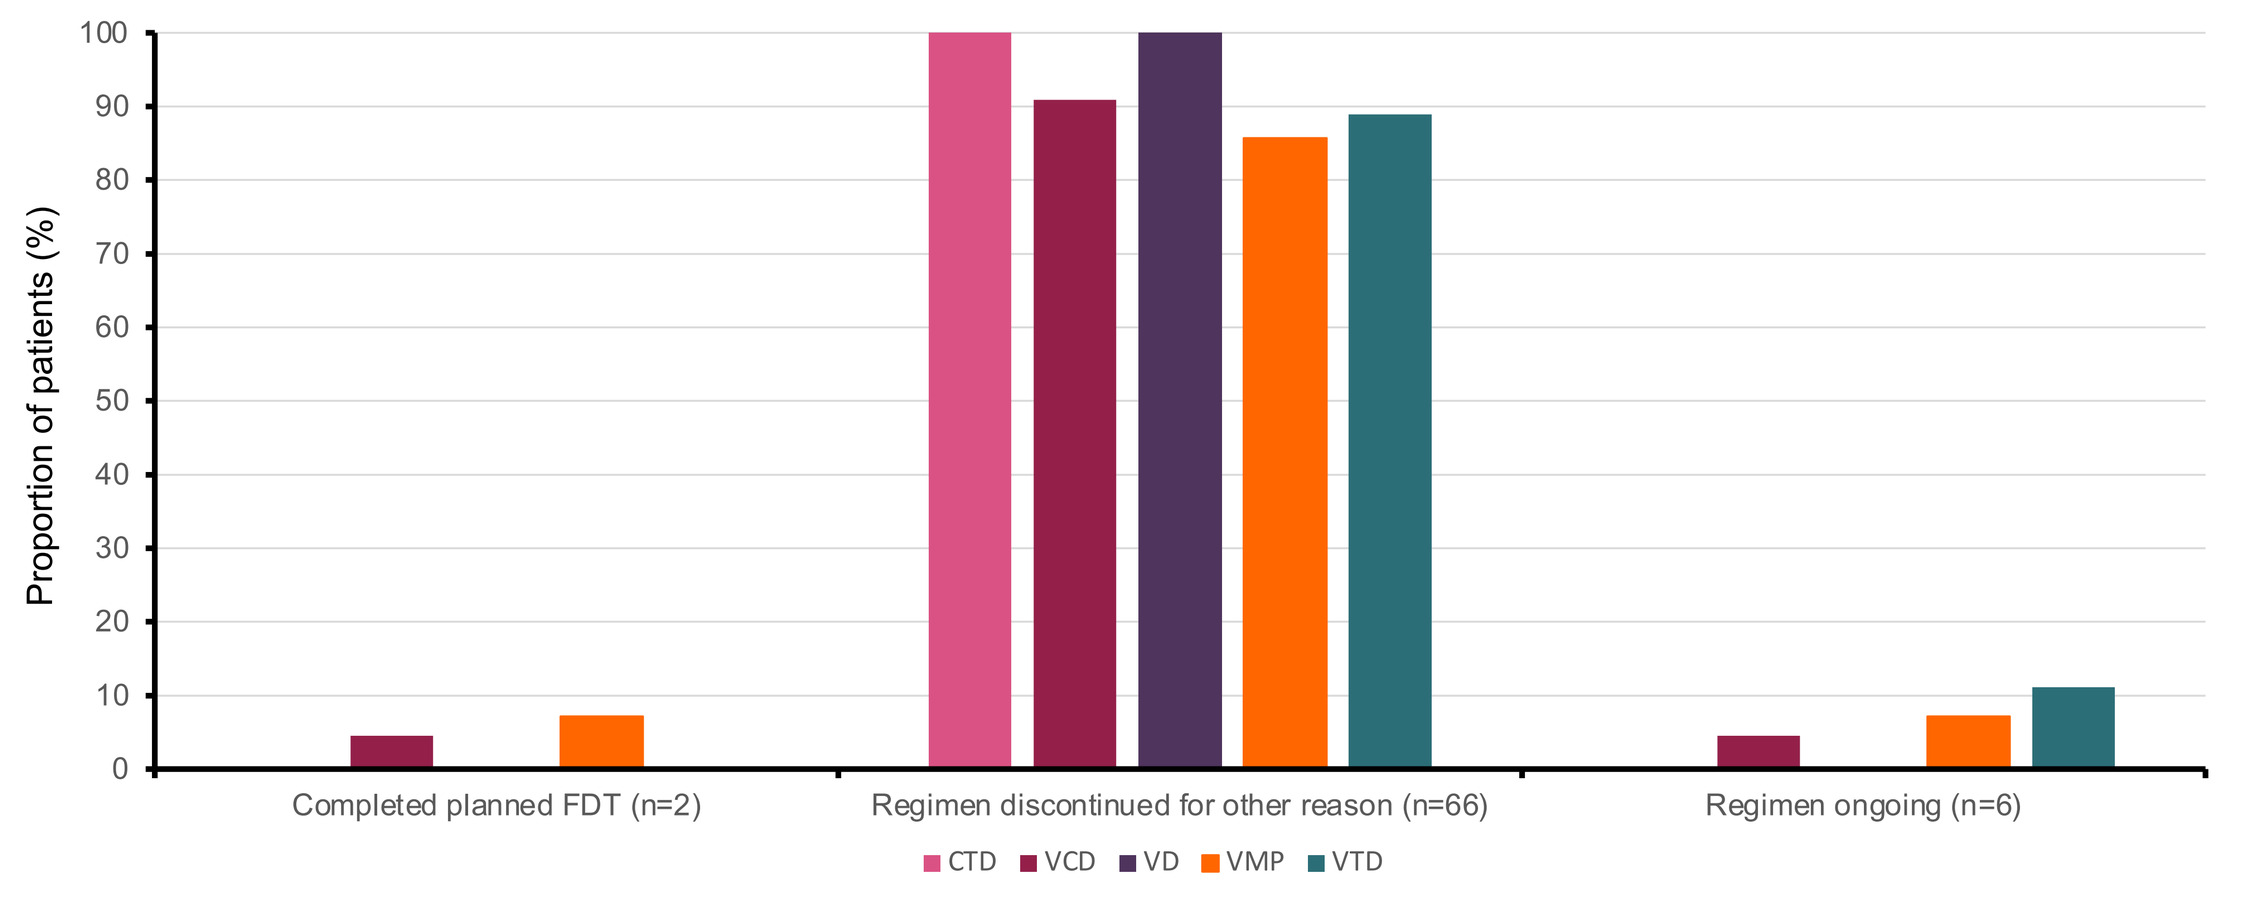
Proportion of patients (%)
Completed planned FDT (n=2)
Regimen discontinued for other reason (n=66)
Regimen ongoing (n=6)
CTD VCD VD VMP VTD

Figure 4c. Completed, discontinued or ongoing therapies – by regimen (LOT2/3)

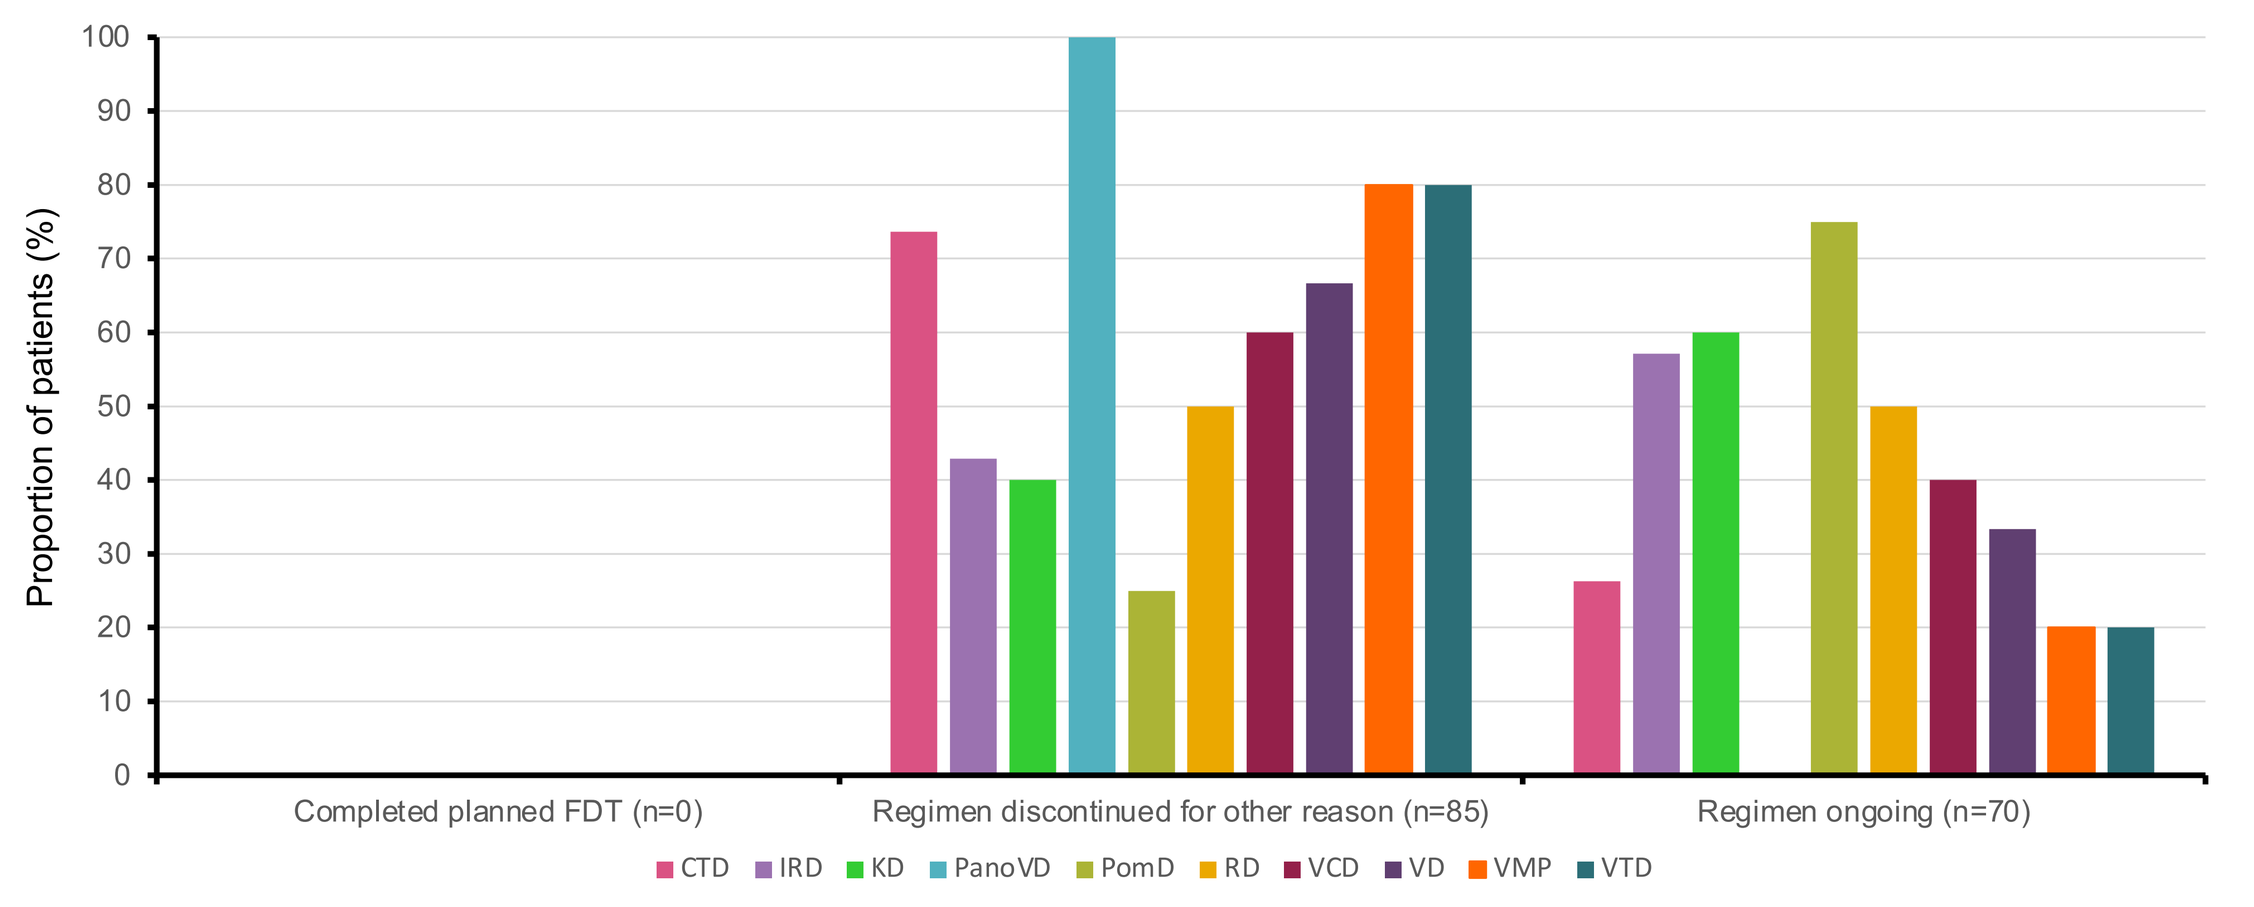
Proportion of patients (%)
Completed planned FDT (n=0)
Regimen discontinued for other reason (n=85)
Regimen ongoing (n=70)
CTD IRD KD PanoVD PomD RD VCD VD VMP VTD

Table 2. DOT by index regimen for combined second and third LOTs

Regimen	Dara (n=6)	IRD (n=27)	KD (n=7)	RD (n=14)	VCD (n=9)	VD (n=9)	VMP (n=4)	VTD (n=7)
Median time to regimen discontinuation, months (95% CI)	NE	16.5 (7.0–NE)	8.1 (1.0–NE)	5.9 (2.2–12.9)	4.4 (1.4–NE)	5.8 (3.2–NE)	8.9 (1.4–NE)	3.9 (1.4–NE)
Time point at which patients still on treatment, n (probability of being on index regimen)								
6 months	4 (0.8333)	14 (0.7622)	3 (0.6429)	4 (0.4571)	1 (0.1905)	3 (0.3889)	2 (0.5)	2 (0.3333)
12 months	1 (0.5556)	6 (0.6987)	0	1 (0.2286)	0	2 (0.3889)	2 (0.5)	1 (0.1667)
18 months	1 (0.5556)	1 (0.4658)		0		0	1 (0.25)	0
24 months	0	0					0	

- Rates of treatment discontinuation due to AEs* (n=22) varied between treatment regimens and where cited, this ranged from 4.2% for PomD to 16.7% for PanoVD (**Figure 5**).
- Relapse was the reason for discontinuation in 19 of 283 patients† with rates, where cited, ranging from 2.1% in patients on VTD to 12.5% in patients on PomD.
- The most common AEs cited as the reason for discontinuation are shown in **Figure 6**.

Figure 5. Reasons for discontinuation – by regimen (any LOT)

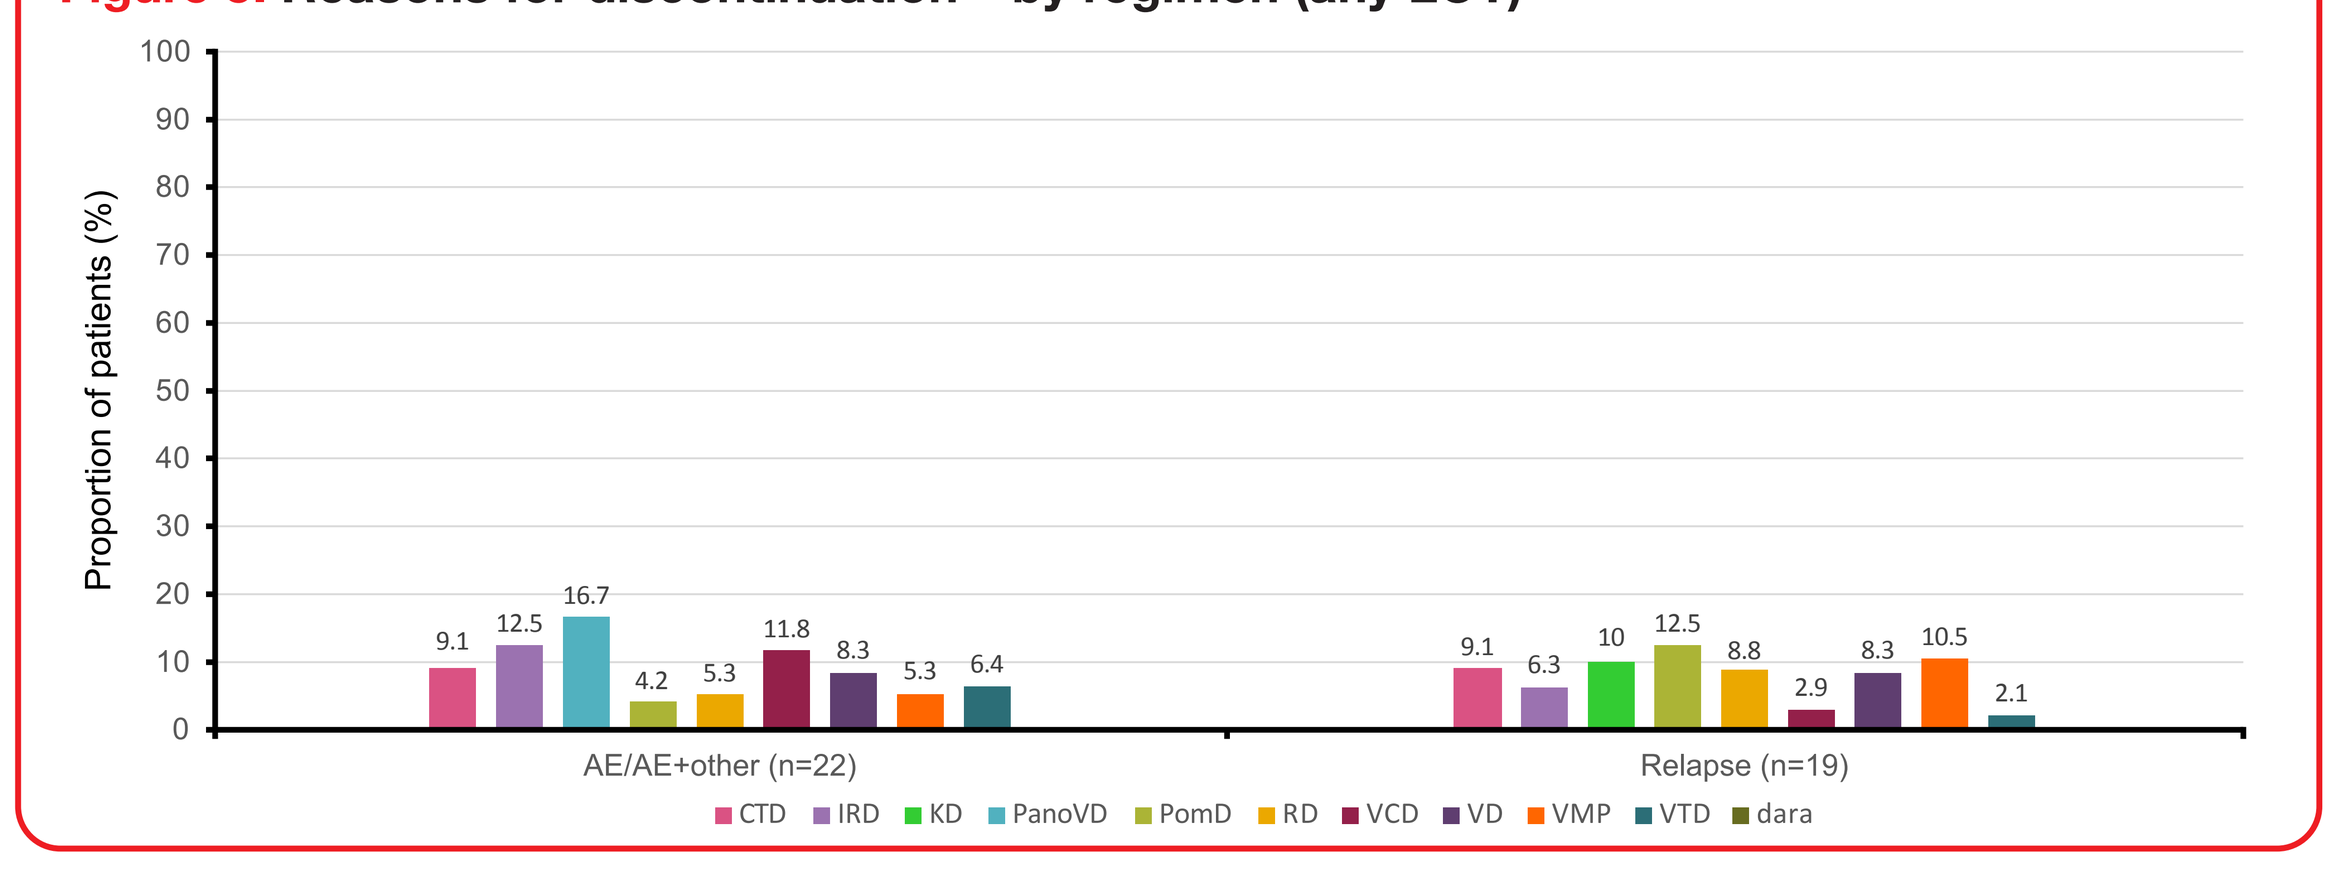
Proportion of patients (%)
9.1 12.5 16.7 4.2 5.3 11.8 8.3 5.3 6.4
9.1 6.3 10 12.5 8.8 2.9 8.3 10.5 2.1
AE/AE+other (n=22)
Relapse (n=19)
CTD IRD KD PanoVD PomD RD VCD VD VMP VTD dara

*Includes reasons given as 'AE' and AE + other'.
†One patient could be counted in different regimens in multiple lines.

Figure 6. Most common AEs leading to discontinuation by line, class and individual drug

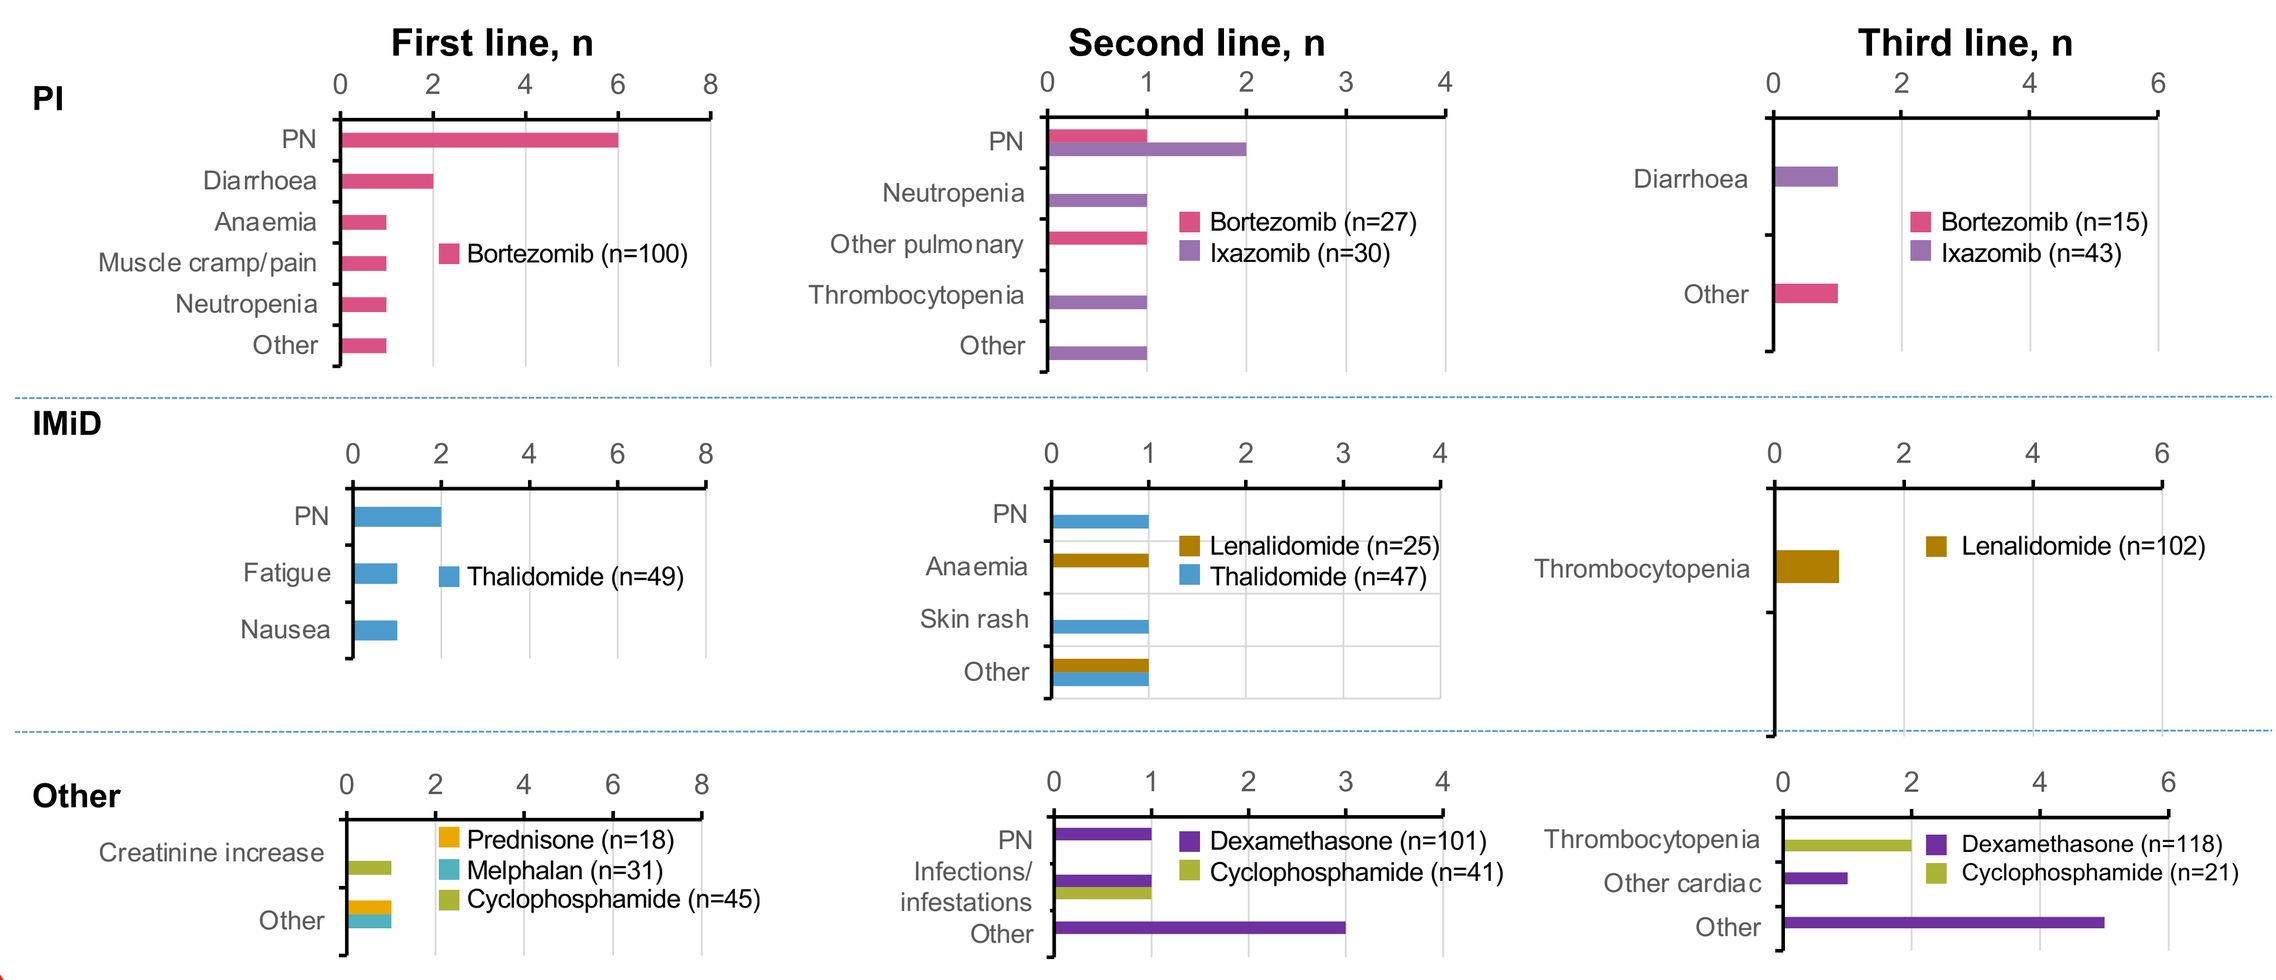
First line, n
Second line, n
Third line, n
PI
PN
Diarrhoea
Anaemia
Muscle cramp/pain
Neutropenia
Other
Bortezomib (n=100)
Neutropenia
Other pulmonary
Thrombocytopenia
Bortezomib (n=27)
Ixazomib (n=30)
Bortezomib (n=15)
Ixazomib (n=43)
IMiD
Fatigue
Nausea
Thalidomide (n=49)
Skin rash
Lenalidomide (n=25)
Thalidomide (n=47)
Lenalidomide (n=102)
Other
Creatinine increase
Prednisone (n=18)
Melphalan (n=31)
Cyclophosphamide (n=45)
Infections/infestations
Dexamethasone (n=101)
Cyclophosphamide (n=41)
Other cardiac
Dexamethasone (n=118)
Cyclophosphamide (n=21)

Discussion

- UK data from INSIGHT MM are consistent with the change in paradigm from fixed duration therapy (FDT) to continuous treatment.
 - Planned end of therapy only accounts for a small proportion of treatment discontinuations, especially in the relapsed setting
 - Patients are discontinuing treatment for reasons other than relapse which means they are ultimately receiving FDT.
- Data from INSIGHT MM support the observation that clinical trial data are not replicated in the real world, with shorter rates of DOT reported in INSIGHT MM than observed in clinical trials.
 - DOT from recent RCTs ranged from 8.9–25.3 months in patients with NDMM[6–10] and 5.0–17.0 months for patients with RRMM.[11–14] The INSIGHT MM UK data shows that shorter DOTs are seen in practice with a range of 3.9–16.5 months for the regimens selected (**Table 2**)
 - Efficacy is broader than survival outcomes from clinical trials and should encompass real-world evidence, quality of life, duration of treatment and safety across agents and lines
 - PN was a common AE, suggesting that it is an impediment to continuous treatment.
- Limitations of the study are the small patient numbers and the need for longer follow up and sourcing of missing data. Given this, and likely treatment variation from centre to centre, more mature data are needed to truly reflect UK-wide practice.
- This analysis of INSIGHT MM shows that there is still no standard of care and no clear pattern for discontinuing treatment and moving to the next LOT.
 - Furthermore, patients are receiving multiple other regimens in addition to the most prescribed regimens.
- More tolerable agents with convenient dosing for continuous treatment and increased access to therapies are needed to improve patient outcomes.

Conclusions

- In this UK-specific analysis from the global INSIGHT MM study, there is clear variability in treatment selection, and reasons for treatment discontinuation change as patients move through lines of therapy.
- The UK INSIGHT MM patient cohort reflects that of the global data; patients are receiving shorter treatment durations than reported in clinical studies.
 - AEs may impact the adherence required to achieve continuous therapy
 - Across lines/agents, PN was a common AE, suggesting that it is an impediment to continuous treatment.
- INSIGHT MM provides a valuable opportunity to understand patterns of MM care in day-to-day clinical practice.

References

1. Palumbo A, et al. J Clin Oncol 2015;33:3459–66.
2. Hungria VTM, et al. Blood 2019;134(Suppl_1):1887.
3. Richardson PG, et al. Blood Cancer J 2018;8:109.
4. Shah JJ, et al. Clin Lymphoma Myeloma Leuk 2017;17:575–83.
5. Costello C, et al. Future Oncol 2019;15:1411–28.
6. Facon T, et al. Blood. 2018;131:301–10.
7. Facon T, et al. N Engl J Med 2019;380(22):2104–15.
8. Facon T, et al. Poster #MM-347, presentation at the virtual 2020 Annual Meeting of SOHO; 9–12 September 2020.
9. Mateos MV, et al. N Engl J Med. 2018;378(6):518–528.
10. Moreau P, et al. Lancet. 2019 Jul 6;394(10192):29–38.
11. Moreau P, et al. N Engl J Med 2016;374:1621–34.
12. Dimopoulos MA, et al. Lancet Oncol 2016;17:27–38.
13. San-Miguel JF et al. Lancet Oncol. 2014;15:1195–1206.
14. Siegel DS et al. Br J Haematol. 2020;188:501–510.
15. Battisti WP, et al. Ann Intern Med 2015;163:461–464.

Abbreviations

AE, adverse event; CI, confidence interval; CTD, cyclophosphamide-thalidomide-dexamethasone; dara, daratumumab; DOT, duration of therapy; EORTC QLQ-C30, European Organization for Research and Treatment of Cancer - Quality of Life Questionnaire – Core 30 module; FDT, fixed duration of therapy; HRQoL, health-related quality of life; HRU, healthcare resource utilisation; IMiD, immunomodulatory drug; IQR, interquartile range; IRD, ixazomib-lenalidomide-dexamethasone; ISS, International Staging System; KD, carfilzomib-dexamethasone; LOT, line of therapy; MM, multiple myeloma; NDMM, newly diagnosed multiple myeloma; NE, not evaluable; PanoVD, panobinostat-bortezomib-dexamethasone; PFS, progression-free survival; PI, proteasome inhibitor; PN, peripheral neuropathy; PomD, pomalidomide-dexamethasone; PRO, patient-reported outcome; QLQ-MY-20, Quality of Life Questionnaire 20-item Multiple Myeloma Module; RCT, randomised controlled trials; RD, lenalidomide-dexamethasone; RRMM, relapsed/refractory multiple myeloma; SCT, stem cell transplantation; SOHO, Society of Hematologic Oncology; TSQM-9, Treatment Satisfaction Questionnaire for Medication-9; VCD, bortezomib-cyclophosphamide-dexamethasone; VD, bortezomib-dexamethasone; VMP, bortezomib-melphalan-prednisone; VTD, bortezomib-thalidomide-dexamethasone.

Disclosures

This study was funded by Takeda Ltd.

GC: Grant/research support from Celgene/BMS, Janssen, and Takeda; Honoraria from Amgen, Bristol-Myers Squibb, Celgene, Janssen, Takeda, Roche and Sanofi. **JA:** Honoraria/educational meeting attendance assistance for Celgene/BMS, Janssen and Takeda. **KB:** Honoraria/educational meeting attendance assistance for Celgene/BMS, Janssen and Takeda. **MG:** Travel grant to attend international meeting/fees for lectures, advisory boards for Amgen, Celgene/BMS, Janssen, Novartis and Takeda. **RH:** Educational support and advisory boards for Celgene/BMA, Janssen, Takeda, and Karyopharm. **BK:** Conference travel grant from Celgene. **SM:** Grants from Janssen; advisory boards for Celgene/BMS, Janssen, Sanofi, Amgen and Takeda; congress sponsorship from Takeda. **DS:** Honoraria from Celgene/BMS, Janssen and Takeda. **NC:** Employee of Takeda. **KR:** Employee of Takeda. **GP:** Advisory boards for/honoraria from Amgen, Beigene, Binding Site, Gilead, Janssen and Takeda.

Acknowledgements

We thank all patients, their families and the investigators at all clinical sites for their participation in the study. We would also like to acknowledge that OPEN Health Medical Communications provided writing support during the development of this poster, which was funded by Takeda Ltd., and complied with Good Publication Practice 3 ethical guidelines.[15]

Poster presentation at the virtual Annual Meeting of the British Society for Haematology (BSH), 9–14 November 2020

BSH2020 PO-124-A
Myeloma
Jill Powell

Poster presented at: British Society for Haematology
Listening • Learning • Leading
PosterSessionOnline.com