HaemBase Cymru (HBC):
An electronic data collection platform for patients treated with haematological malignancies in Wales.

Ceri Bygrave*[1], Zeyad Khalaf[2], Liz Edwards[2]
[1]Haematology, University Hospital of Wales, Cardiff, [2]Haematology, Janssen-Cilag, High Wycombe, United Kingdom. Medical writing support provided by Matthew Wilson, Janssen, UK

This is a wonderful development for cancer services in Wales. What is being developed in myeloma will soon be available for all haemato-oncology patients, and will be the blueprint for solid tumour cancer services in the future too. This is exactly what clinicians have been waiting for and will hugely support communications between all professionals across the patient pathway.

- Prof. Tom Crosby OBE, Consultant Oncologist, National Cancer Clinical Director for Wales, Clinical Lead Transforming Cancer Services.

Background & Objectives

HCB is an innovative partnership between NHS Wales, the Wales Cancer Network, Welsh Government and Janssen to drive improved data capture and patient-centred care for haematological malignancies. This five-year joint working agreement began in Jan 2018 between the four parties where this solution has been designed and developed by NWIS (NHS Wales Informatics Service) as part of the national data architecture confirming complete alignment with existing and future national systems. Welsh Government (WG) and Public Health Wales (PHW) are committed to ensuring the provision of sustainable services through NHS Wales' National Planned Care Programme.

Aims

The aim of this joint working agreement includes putting Wales at the forefront of measuring and tracking patient outcomes, using new technology in the form of a purpose- built haematological malignancy data solution, with the aim of making innovative new treatments available in line with the aspirations in the Cancer Delivery Plan.

Methods & Results

Working with a wide stakeholder group of around 40 members across Wales including clinicians, service users, Myeloma UK, Cancer Network members and government agencies HBC has culminated in the development of an e-form (Outpatient Continuation Sheet, OCS Myeloma) which can be accessed by any clinician via the Welsh Clinical Portal (existing national system). The system will be capturing real time data on all myeloma patients in Wales. It can capture clinical data, MDT decisions, treatment, responses and quality of life data. This facilitates robust analysis of the landscape of the disease, the impact of treatment and overall patient benefit through comprehensive capture of real-world population data. This supports prudent use of resources including high costs anti-cancer drugs.

HBC went live in Dec 2019 in Cardiff & Vale Health Board at the Multiple Myeloma clinic of Dr Ceri Bygrave who is the lead clinician within HBC. Dr Bygrave has now been using HBC for new and follow up patients in Cardiff capturing patient level data in real time. It is the first system in the UK, purpose built for myeloma patients initially, leading to all patients with haematological malignancies, with the aim of creating a single electronic point of data entry via a national NHS portal. The solution is also the trailblazer for the wider Cancer Informatics Solution which has secured government funding and will be developed and built by NWIS.

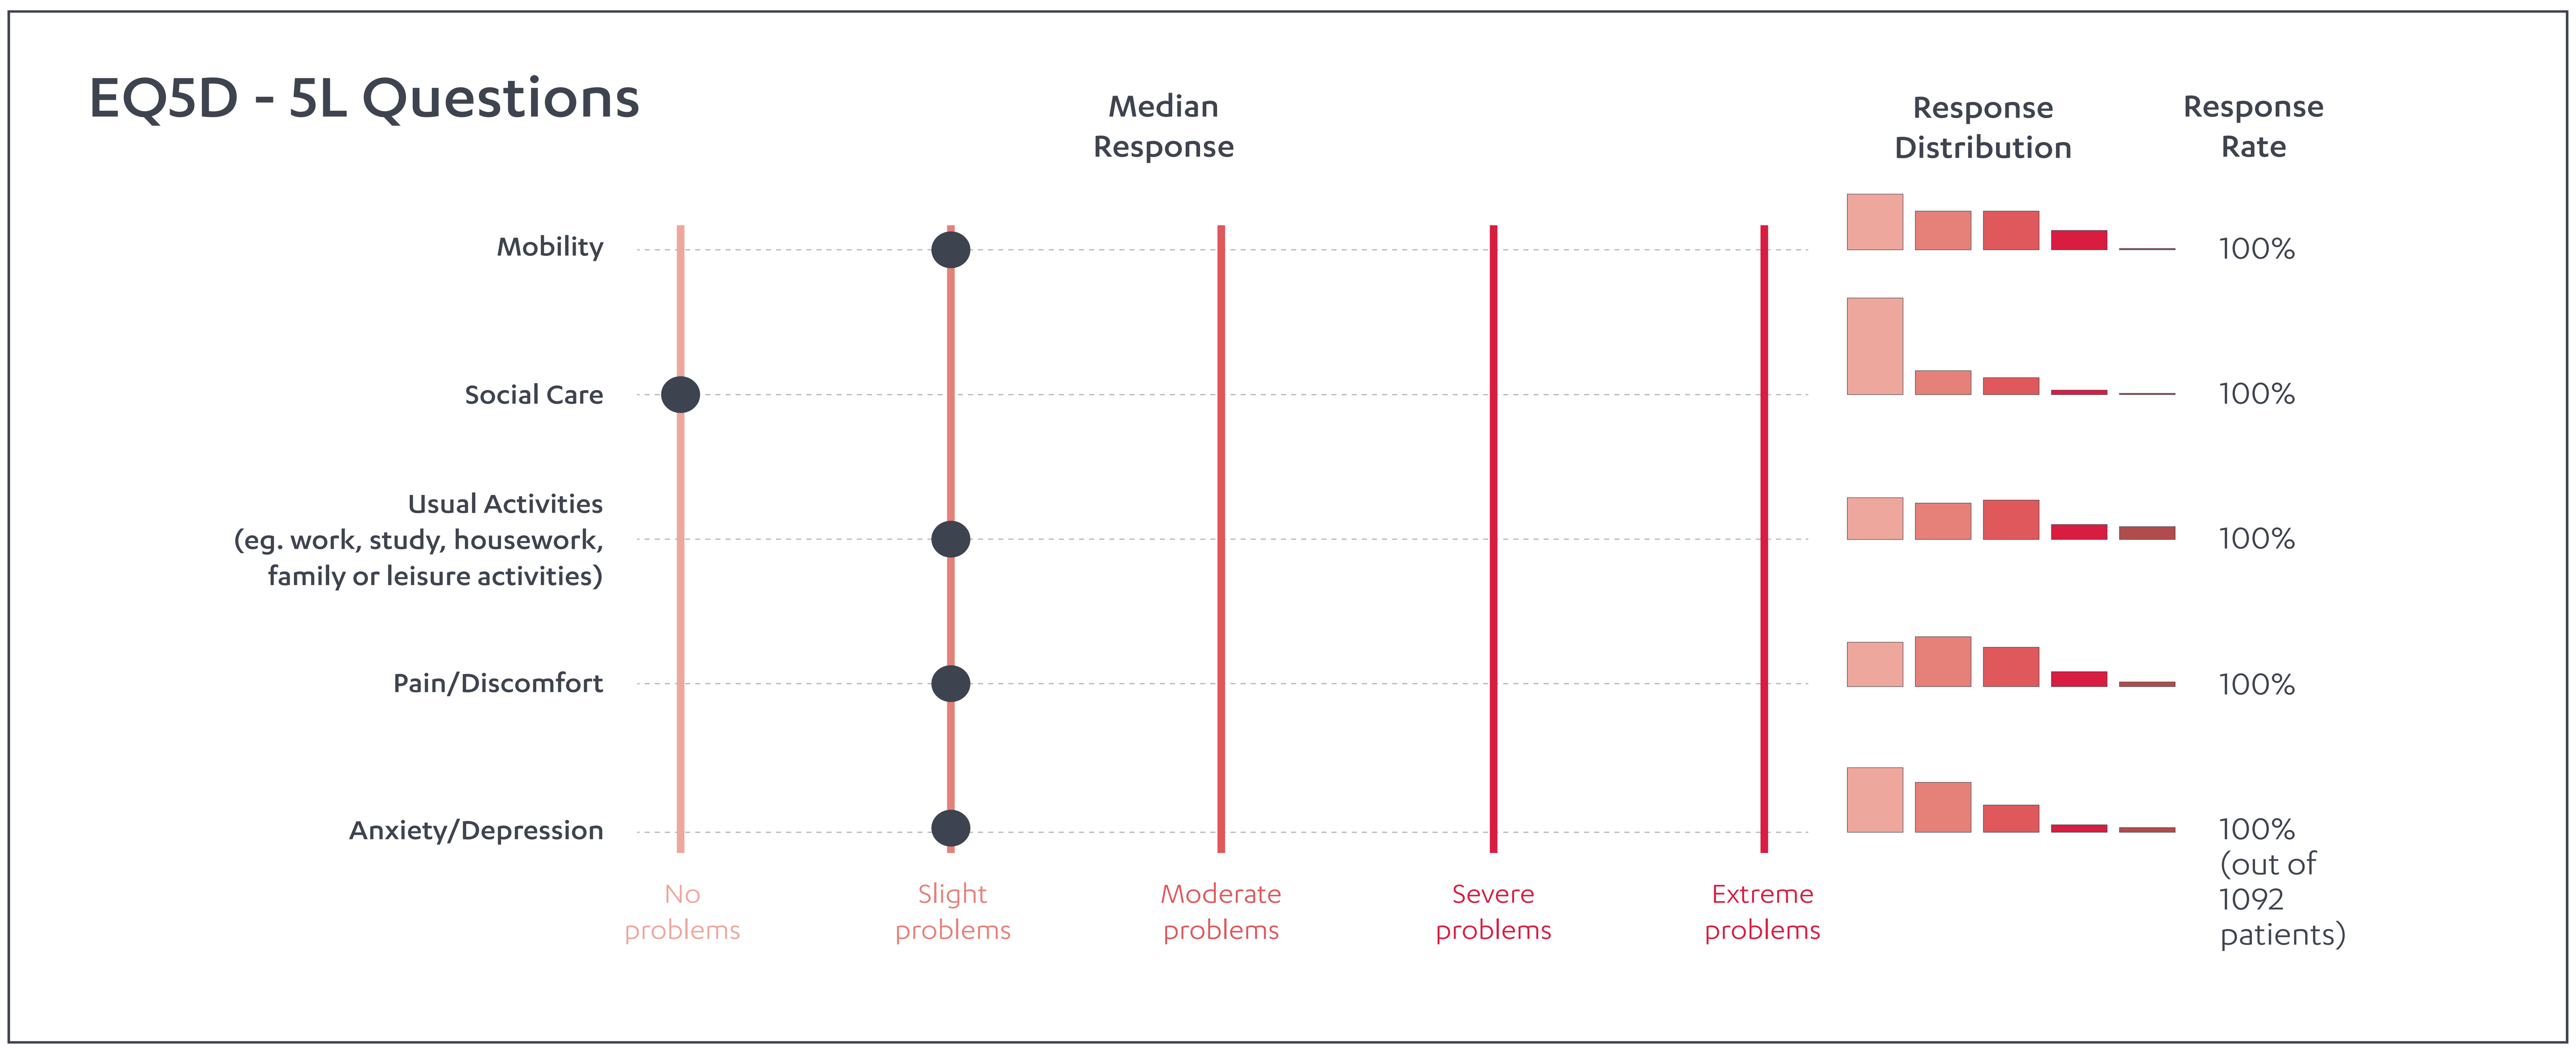
EQ5D - 5L Questions
Median Response
Response Distribution
Response Rate
Mobility
Social Care
Usual Activities (eg. work, study, housework, family or leisure activities)
Pain/Discomfort
Anxiety/Depression
No problems
Slight problems
Moderate problems
Severe problems
Extreme problems
100%
100%
100%
100%
100% (out of 1092 patients)

Initially this pilot, which ran for 3 months and recruited 1092 patients, has focused on multiple myeloma, and the collection of Patient Reported Outcome Measures (PROMs) in this patient group has been identified as a key indicator. The MyPOS (Myeloma-specific Palliative care Outcome Score) was chosen as an appropriate tool, to be collected alongside the generic EQ5D-5L, and a pilot has been undertaken to assess the feasibility of collecting PROMs in this patient group. This pilot was undertaken within Aneurin Bevan Health Board, and myeloma patients were invited to complete the MyPOS and EQ5D PROMs via the DrDoctor electronic platform.

Summary

Patients with haematological malignancies have a clear and concise pathway, with clinicians able to digitally access records from any clinical setting in real time. Clinicians will be able to make informed treatment decisions based on accurate data capture. PROMS will be captured on the national platform and integrated into HBC, to provide a comprehensive patient-level data set that can inform patient/clinician interactions and support assessment of value in the pathway. This will provide a 360-degree view of the individual patient's journey at a single point and will also generate quality of life and clinical outcome data for the entire Welsh population. As a result, NHS Wales will have meaningful analysis of the impact of treatment and support prudent use of healthcare resources.

Acknowledges: Janssen has partnered with Welsh Government, Public Health Wales and Cardiff Vale University Health board on a joint working initiative to create a personalised database that can critically analyse the outcome data for patients with myeloma - for more information, please visit: https://www.janssen.com/uk/our-partnerships/working-with-our-partners. NHS Wales Informatics Service (NWIS), Welsh Government, Wales Cancer Network and NHS Wales who have been instrumental in the conception, evolution and delivery of this platform.

EM-42222 September 2020

12th Dec 2019

Dr Ceri Bygrave and team at UHW commenced an Early Adopter evaluation of the first part of the solution, i.e. the Outpatient Continuation Sheet (Myeloma Iteration).

After the initial evaluation, further iterations of the OCS will be released to cater for all Haematological Malignancies and to be rolled out for adoption in other Haematology Clinics.

Jan - June 2020

Other Health Boards will start to implement solution supported by e-learning, NWIS and Haem Base CymruProject members.

A first iteration of functionalities to be used in the Multi Disciplinary Team (MDT) meetings is also scheduled to be developed in this timeframe and will be the focus of the first 9 months of 2020.

Please contact Elizabeth Edwards (eedward7@ITS.JNJ.com) for further information about HaemBaseCymru.

BSH2020
EP-107-A
Myeloma
Zeyad Khalaf
Poster presented at: British Society for Haematology
Listening • Learning • Leading
PosterSessionOnline.com