SP428

A randomized, single-center, double-blind, placebo-controlled, multiple-dose, phase 1 study to evaluate the safety, pharmacokinetics, and pharmacodynamics of TAK-272 in healthy adult non-elderly and elderly male subjects

Kumi Matsuno, Shingo Tanaka, Takamasa Hashimoto, Hiroyuki Nakamichi, Emiko Komura

Takeda Pharmaceutical Company Ltd., Osaka, Japan

Introduction

- Renin is a rate-limiting enzyme that initiates the enzymatic cascade of the renin-angiotensin system
- The successful development of the direct renin inhibitor (DRI) aliskiren for hypertension validated direct renin inhibition as a therapeutic strategy for cardiovascular/renal disease
 - However, the bioavailability of aliskiren is low and the maximum dosage is limited by gastrointestinal toxicity
- TAK-272 is a novel, potent, and orally active DRI that has demonstrated good bioavailability and stronger in vivo renin inhibition compared with aliskiren in preclinical animal studies[1]
- In a single-dose study conducted in Japan (CPH-001), TAK-272 at a dose up to 200 mg demonstrated good tolerability in healthy adult male subjects[2]
- Based on the results of study CPH-001, we evaluated the safety, pharmacokinetics (PK), and pharmacodynamics (PD) of multiple oral administrations of TAK-272 at doses of 80 mg and 160 mg in healthy adult non-elderly and elderly male subjects

Methods

- This study (CPH-002) was a randomized, placebo-controlled, double-blind, multiple-dose phase 1 study, comprising 3 cohorts evaluated consecutively (Table 1)
 - Subjects were randomized within 1 of the 3 cohorts to receive TAK-272 or placebo
- Primary objective: evaluate the safety and the PK of multiple oral administration of TAK-272 tablets in healthy Japanese adult non-elderly and elderly male subjects
 - Secondary objective: evaluate the PD of multiple oral administration of TAK-272 tablets in healthy Japanese adult non-elderly and elderly male subjects
- Elderly (age 65–85 years) or non-elderly (age 20–45 years) healthy Japanese male subjects with weight ≥50 kg and body mass index of ≥18.5 kg/m^2 and ≤25.0 kg/m^2 were eligible
- TAK-272 or placebo tablets were administered orally, once daily, for 7 days
 - Subjects received the study drug 30 minutes after breakfast with 200 mL of water
- Food and drink containing grapefruit, caffeine, and alcohol were prohibited from 72 hours prior to the start of administration through discharge
 - There were no other dietary restrictions in the study, including restrictions on dietary salt
- Safety evaluations included adverse events, vital signs, weight, electrocardiogram (ECG), and laboratory tests
- PK and PD evaluations:
 - Plasma and urinary concentrations of TAK-272F and its metabolite, M-I, were measured by high-performance liquid chromatography-tandem mass spectrometry
 - Plasma renin activity (PRA), angiotensin I concentration, AII concentration, and plasma aldosterone concentration were measured by radioimmunoassay. Plasma renin concentration (PRC) was measured by immunoradiometric assay
- PK parameters were estimated from plasma concentration-time profiles for each analyte using WinNonlin® (Pharsight Corporation, a Certara Company, Mountain View, CA) by non-compartmental analysis

Table 1. Dose and number of subjects[a]

		Number of subjects	
Cohort	**Dose**	**TAK-272**	**Placebo**
1 (non-elderly)	TAK-272 80 mg or placebo	9	3
2 (non-elderly)	TAK-272 160 mg or placebo	9	3
3 (elderly)	TAK-272 80 mg or placebo	9	3

[a]The study started with the evaluation of Cohort 1. After comprehensive review of the safety results of Cohort 1 up to 168 hours after the last study drug administration, evaluation of Cohort 2 was initiated. After comprehensive review of the safety results of Cohorts 1 and 2 up to 168 hours after the last study drug administration, evaluation of Cohort 3 was initiated. The interval between the last study drug administration of one cohort and the first study drug administration of the subsequent cohort was ≥8 days.

Results

Patient Disposition

- A total of 78 subjects were screened and 46 were enrolled in the study (Figure 1)
- Thirty-six subjects were randomized and 35 completed the study
 - One subject was withdrawn from the study after using excluded concomitant medications
- The safety, PK, and PD analysis sets each included all 36 subjects who received study drug

Figure 1. Patient disposition

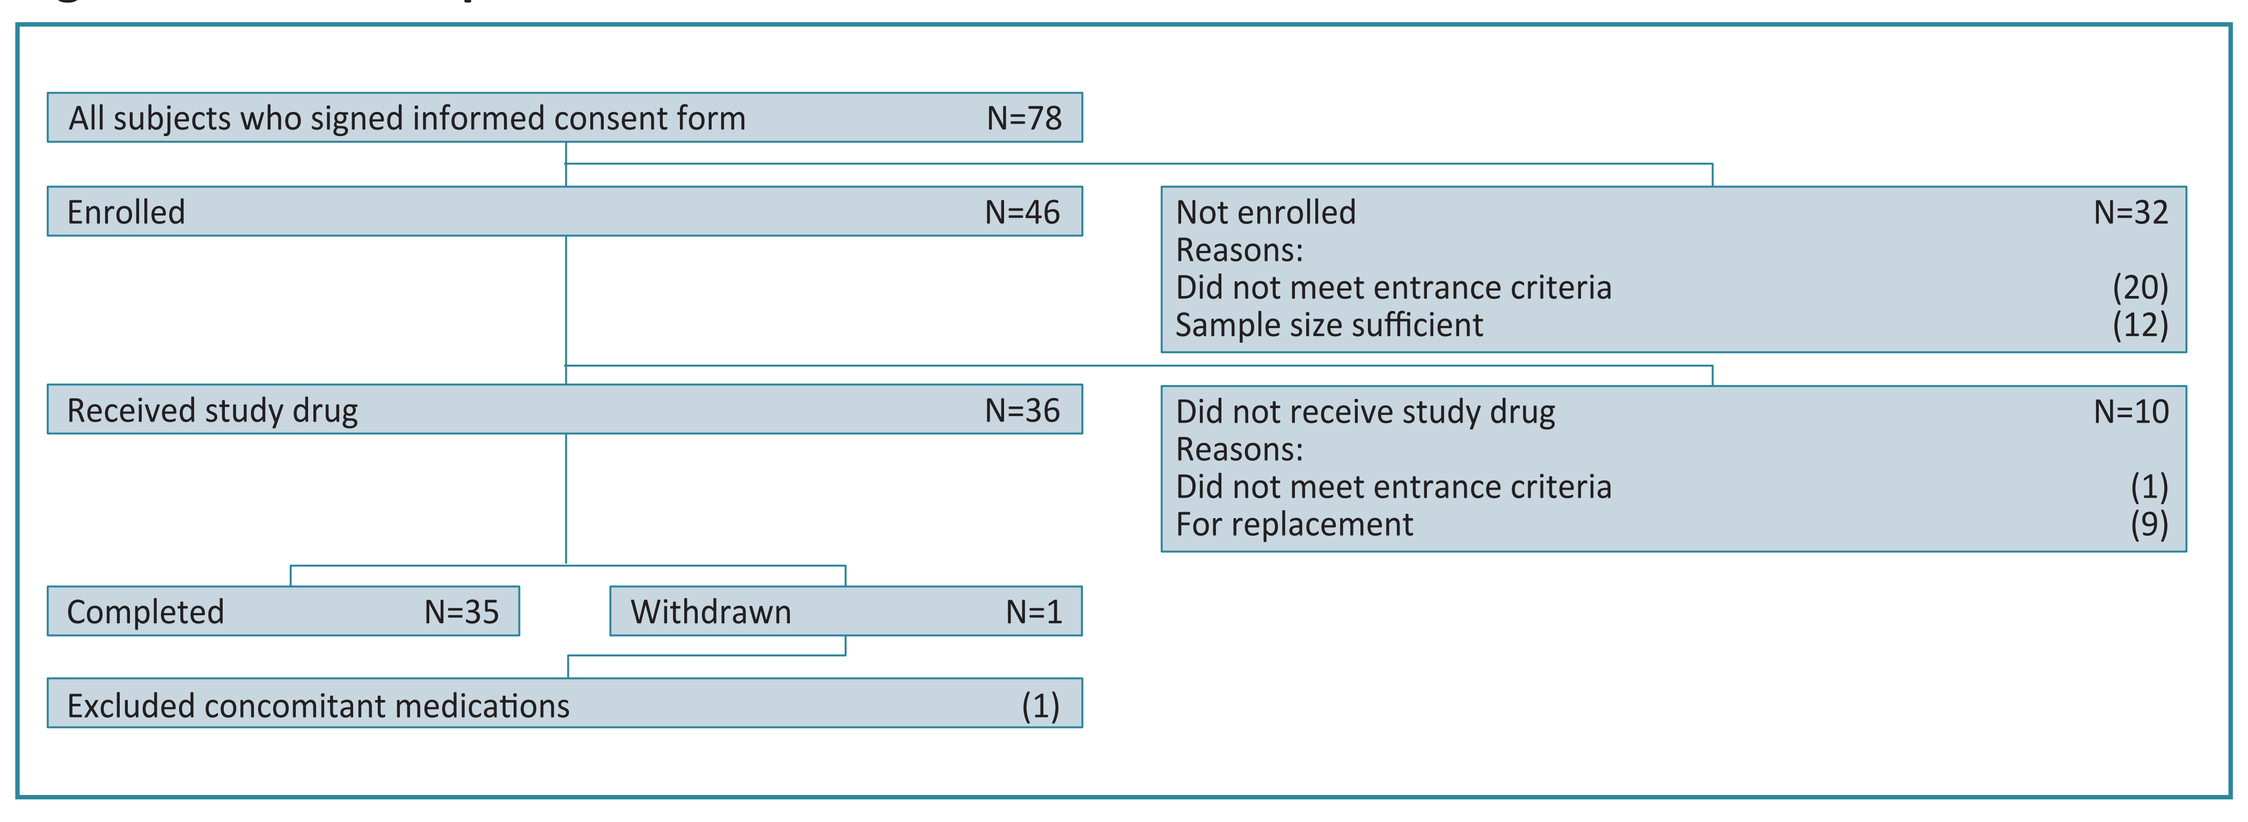
All subjects who signed informed consent form N=78
Enrolled N=46
Not enrolled N=32
Reasons:
Did not meet entrance criteria (20)
Sample size sufficient (12)
Received study drug N=36
Did not receive study drug N=10
Reasons:
Did not meet entrance criteria (1)
For replacement (9)
Completed N=35
Withdrawn N=1
Excluded concomitant medications (1)

Baseline Subject Characteristics

- Baseline characterists are shown in Table 2
- The mean creatinine clearances of non-elderly groups ranged from 136.4 mL/min to 143.7 mL/min, and the mean creatinine clearances in elderly groups ranged from 74.2 mL/min to 77.3 mL/min
- There were no obvious differences in demographic and baseline characteristics between the TAK-272 and placebo groups within both non-elderly and elderly cohorts

Table 2. Baseline characteristics

	Non-elderly[a]			Elderly[b]	
	Placebo (n = 6)	**TAK-272 80 mg (n = 9)**	**TAK-272 160 mg (n = 9)**	**Placebo (n = 3)**	**TAK-272 80 mg (n = 9)**
Age (years), mean (SD)	26.7 (6.5)	26.4 (5.5)	24.8 (3.8)	72.7 (4.0)	72.1 (5.4)
Height (cm), mean (SD)	171.7 (4.1)	169.0 (5.3)	174.3 (6.2)	165.7 (2.9)	163.0 (7.6)
Weight (kg), mean (SD)	67.5 (4.6)	61.9 (6.8)	63.2 (8.0)	60.7 (2.2)	59.1 (8.2)
BMI (kg/m^2), mean (SD)	22.9 (1.6)	21.7 (2.5)	20.8 (2.1)	22.2 (1.2)	22.2 (1.7)
AGP (mg/dL), mean (SD)	58.2 (15.0)	59.6 (8.2)	55.0 (9.8)	41.1 (5.7)	62.2 (12.5)
Creatinine clearance (mL/min), mean (SD)	143.7 (19.5)	136.4 (11.1)	140.8 (24.7)	77.3 (14.5)	74.2 (17.0)
Smoking classification, n (%)					
Never smoked	4 (66.7)	6 (66.7)	4 (44.4)	3 (100)	8 (88.9)
Current smoker	1 (16.7)	1 (11.1)	2 (22.2)	0	0
Ex-smoker	1 (16.7)	2 (22.2)	3 (33.3)	0	1 (11.1)
PRA (ng/mL/hr), mean (SD)	1.3 (1.1)	3.1 (2.3)	1.1 (1.0)	2.0 (1.0)	1.6 (0.7)
PRC (pg/mL), mean (SD)	7.1 (4.8)	11.7 (7.1)	6.2 (3.5)	4.3 (0.3)	5.3 (2.0)

[a]Subjects aged 20 to 45 years, inclusive.
[b]Subjects aged 65 to 85 years, inclusive.
AGP, alpha-1 acid glycoprotein; BMI, body mass index; PRA, plasma renin activity; PRC, plasma renin concentration; SD, standard deviation.

Results (continued)

Pharmacokinetics

Non-elderly subjects

- Descriptive statistics for PK parameters of TAK-272F are shown in Table 3
- Based on the trough concentration of TAK-272F, the plasma concentration of TAK-272F reached the steady state by Day 7 during multiple oral administration of 80 mg and 160 mg tablets (Figure 2A)
- Accumulation factors and indices are shown in Table 4
- TAK-272F was the main compound detected in plasma; at any dose, the $AUC_{0–tau}$ of M-I on Day 7 was <3% that of TAK-272F
- Cumulative urinary excretion percentages (% of TAK-272 dose) of TAK-272F up to 72 hours after the last dose of TAK-272 were 11.9% and 12.9% for the 80 and 160 mg doses, respectively

Table 3. PK parameters of TAK-272F following multiple oral administrations of TAK-272 in non-elderly and elderly subjects

	Day 1			Day 7		
	TAK-272 80 mg		TAK-272 160 mg	TAK-272 80 mg		TAK-272 160 mg
Parameters	**Non-elderly (n = 9)**	**Elderly (n = 9)**	**Non-elderly (n = 9)**	**Non-elderly (n = 9)**	**Elderly (n = 9)**	**Non-elderly (n = 9)**
AUC_{0-tau} (ng*hr/mL), mean (SD)	3732 (780.4)	4938 (1795.3)	8008 (2026.8)	3937 (991.4)	6017 (1701.9)	9528 (2570.3)
AUC_{0-inf} (ng*hr/mL), mean (SD)	4217 (863.8)	5886 (2282.8)	8599 (2108.0)	—	—	—
C_{max} (ng/mL), mean (SD)	764.3 (275.4)	958.9 (412.2)	2021 (819.1)	754.2 (255.3)	1071 (520.2)	1859 (417.1)
T_{max} (hr), median (min–max)	2.0 (1.0–3.0)	1.5 (1.0–4.0)	1.0 (1.0–2.5)	2.0 (1.0–3.0)	2.0 (1.0–4.0)	1.5 (1.0–3.0)
$T_{1/2}$ (hr), mean (SD)	10.3 (4.5)	11.2 (1.5)	8.0 (1.0)	11.0 (2.8)	14.8 (3.1)	9.4 (2.2)
CL/F (L/hr), mean (SD)	19.6 (3.6)	15.2 (4.6)	19.7 (5.1)	21.5 (5.6)	14.1 (3.4)	17.6 (3.3)

AUC_{0-tau}, area under the plasma concentration-time curve from time 0 to time tau; AUC_{0-inf}, area under the plasma-concentration time curve from time 0 to infinity; CL/F, apparent total clearance; C_{max}, maximum observed plasma concentration; SD, standard deviation; TAK-272F, unchanged TAK-272; T_{max}, time to reach the maximum observed plasma concentration; $T_{1/2}$, apparent elimination half-life in the terminal elimination phase.

Table 4. Accumulation factor and accumulation index of TAK-272F following multiple oral administrations of TAK-272 in non-elderly subjects

Parameters	**TAK-272 80 mg (n = 9)**	**TAK-272 160 mg (n = 9)**
Accumulation factor (AUC)[a]	1.1 (0.2)	1.2 (0.3)
Accumulation factor (C_{max})[b]	1.0 (0.4)	1.0 (0.3)
Accumulation index (AUC)[c]	0.9 (0.2)	1.1 (0.3)
Accumulation index ($T_{1/2}$)[d]	1.2 (0.4)	1.2 (0.3)

[a]Defined as AUC_{0-tau} on Day 7/AUC_{0-tau} on Day 1.
[b]Defined as C_{max} on Day 7/C_{max} on Day 1.
[c]Defined as AUC_{0-tau} on Day 7/AUC_{0-inf} on Day 1.
[d]Defined as $T_{1/2}$ on Day 7/$T_{1/2}$ on Day 1.
AUC, area under the plasma concentration-time curve, C_{max}, maximum observed plasma concentration; $T_{1/2}$, apparent elimination half-life in the terminal elimination phase; TAK-272F, unchanged TAK-272.

Figure 2. (A) Arithmetic mean time profile of plasma concentrations of TAK-272F following multiple oral administrations of TAK-272 (80 or 160 mg) in non-elderly subjects. (B) Arithmetic mean time profile of plasma concentrations of TAK-272F following multiple oral administrations of TAK-272 80 mg in non-elderly and elderly subjects

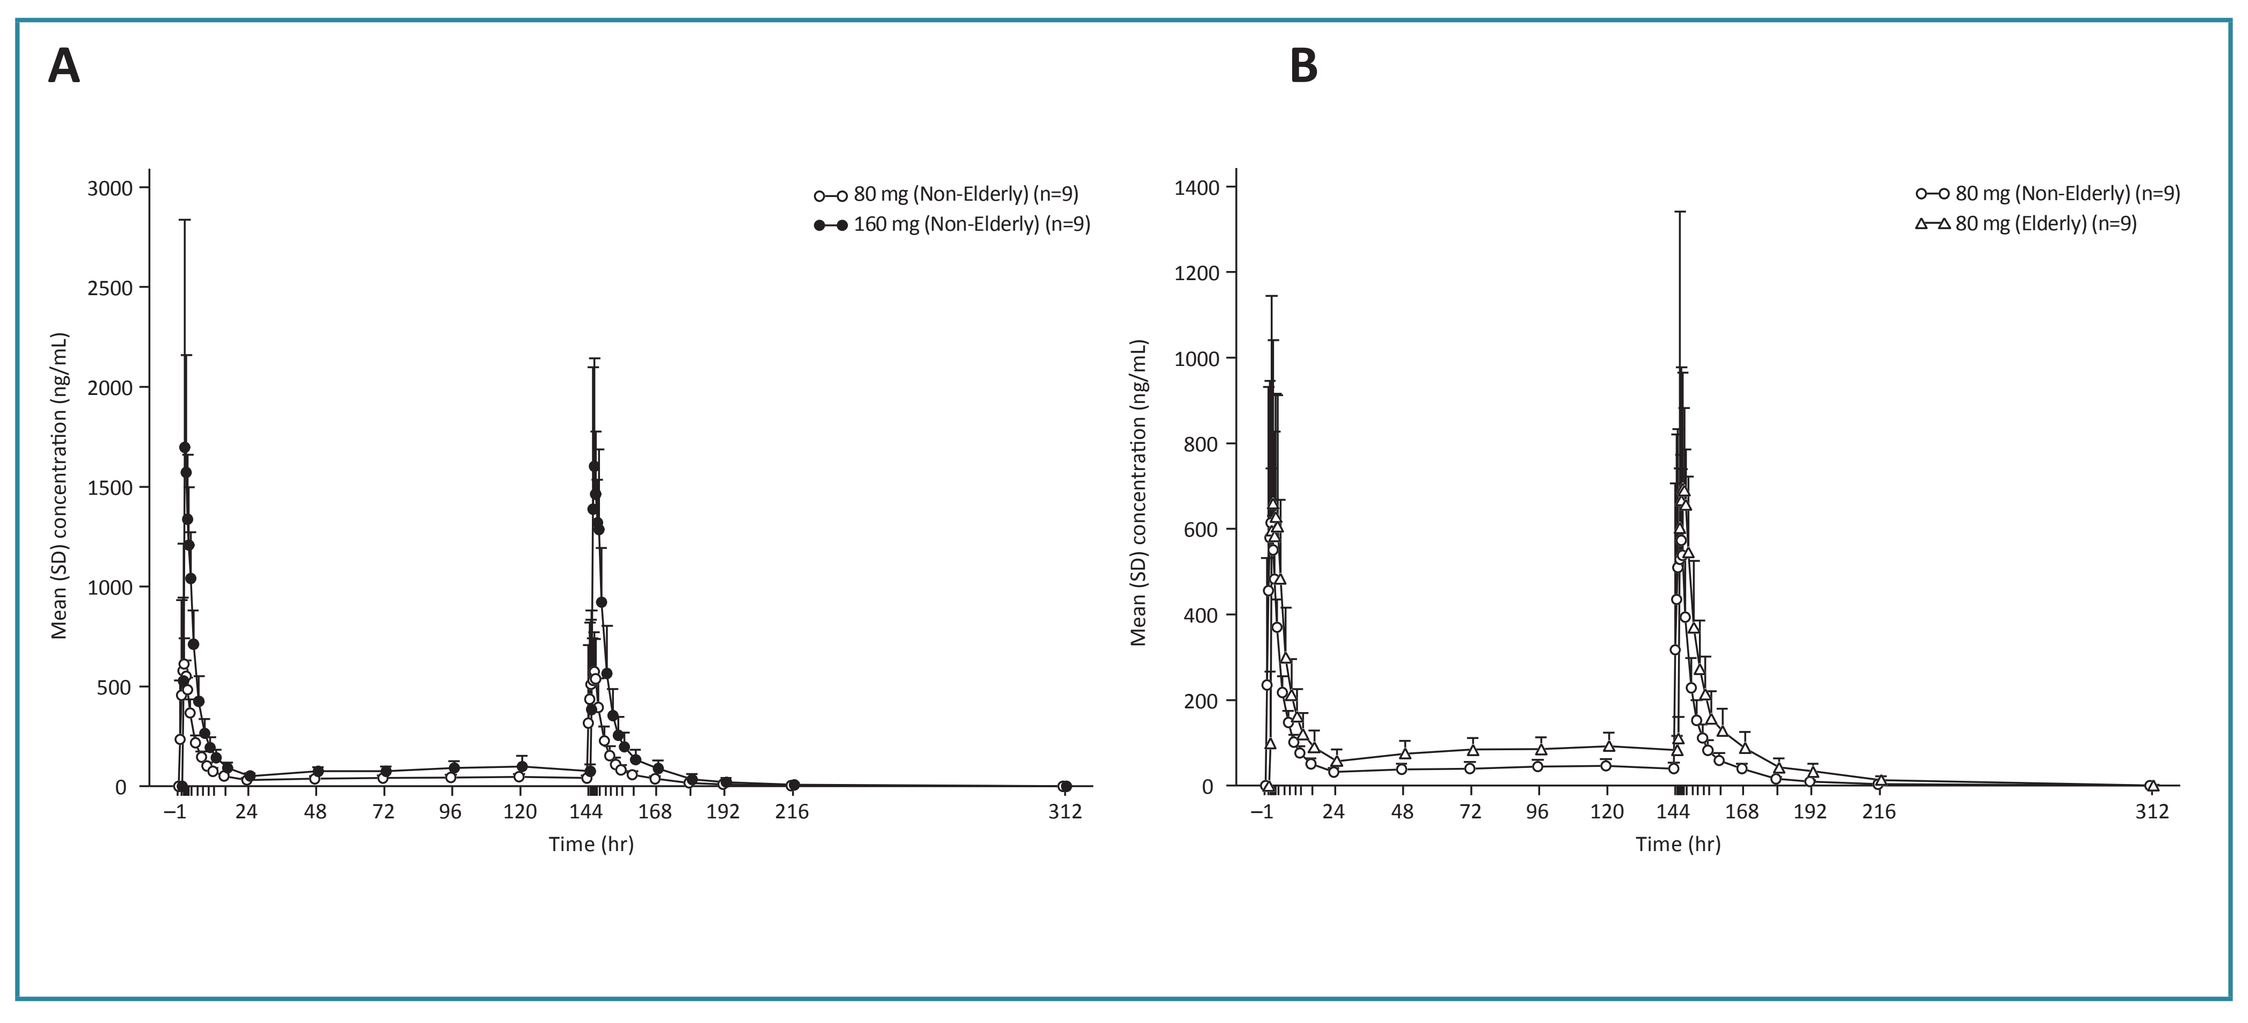
A
B
80 mg (Non-Elderly) (n=9)
160 mg (Non-Elderly) (n=9)
80 mg (Elderly) (n=9)
Mean (SD) concentration (ng/mL)
Time (hr)

SD, standard deviation; TAK-272F, unchanged TAK-272.

Age effect on PK of TAK-272F

- Arithmetic mean time profiles of the plasma concentration of TAK-272F following multiple oral administration of 80 mg TAK-272 in non-elderly and elderly subjects are shown in Figure 2B
- The mean $T_{1/2}$ of TAK-272F and M-I were longer in elderly subjects than in non-elderly subjects on Day 7
- $AUC_{0–tau}$ and C_{max} of TAK-272F were higher in elderly than in non-elderly subjects (52% and 39% higher, respectively)
 - The AUC and C_{max} of M-I were greater in elderly subjects than in non-elderly subjects on Day 1 and Day 7
- Cumulative urinary excretion percentage (% of TAK-272 dose) of TAK-272F up to 72 hours after the last dose of TAK-272 was 13.8% in elderly subjects

Pharmacodynamics

Non-elderly subjects

- The PRA was rapidly inhibited at any dose in non-elderly subjects on Day 1. The strong inhibition of PRA was observed for 7 days, and maintained until 71 hours after the last administration of TAK-272 (Figure 3A)
- The PRC increased at any dose, and the increase from baseline was observed for 7 days (Figure 3B)
 - The peak concentrations of PRC were at similar levels across doses on Day 1
 - However, the peak concentrations of PRC in the TAK-272 160-mg group was higher on Day 7 than those in the TAK-272 80-mg group
 - In both dose groups, the peak concentrations of PRC were higher on Day 7 than on Day 1
 - Trough PRC continued to increase for at least 47 hours postdose

Figure 3. Arithmetic mean time profiles of inhibition rate of PRA (A) and change from baseline in PRC (B) following multiple oral administrations of TAK-272 (80 or 160 mg) in non-elderly subjects

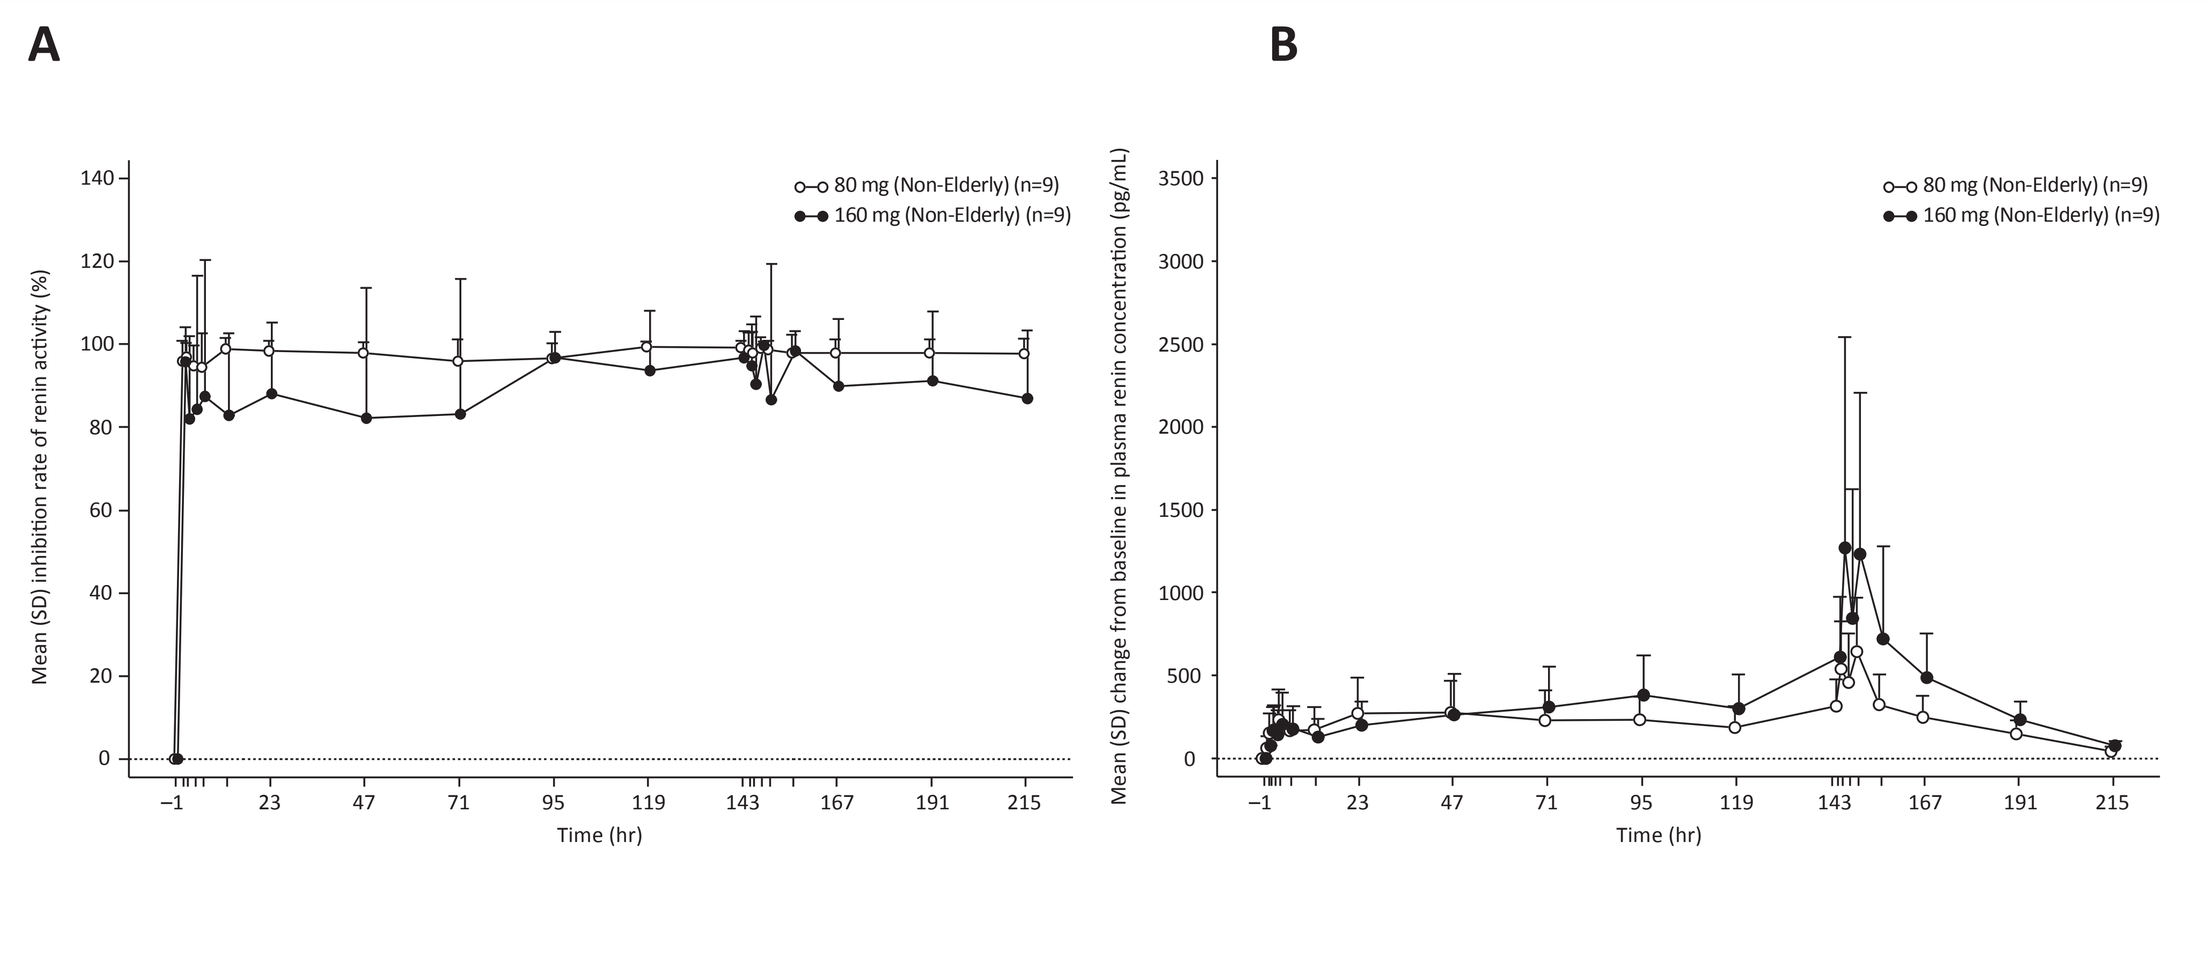
A
B
80 mg (Non-Elderly) (n=9)
160 mg (Non-Elderly) (n=9)
Mean (SD) inhibition rate of renin activity (%)
Mean (SD) change from baseline in plasma renin concentration (pg/mL)
Time (hr)

PRA, plasma renin activity; PRC, plasma renin concentration; SD, standard deviation.

Age effect on PD

- The strong inhibition of PRA was also observed in elderly subjects for 7 days as with non-elderly subjects, and maintained until 71 hours after the last administration of TAK-272 (Figure 4A)
- The PRC increased in elderly subjects, and the increase from baseline was observed for 7 days
 - The peak concentrations of PRC were higher on Day 7 than on Day 1 as with in non-elderly subjects
 - Increase from baseline in PRC was smaller in elderly subjects than in non-elderly subjects during the 7 days of treatment period and until 71 hours after the last administration of TAK-272 (Figure 4B)

Figure 4. Arithmetic mean time profiles of inhibition rate of PRA (A) and change from baseline in PRC (B) following multiple oral administrations of TAK-272 (80 mg) in non-elderly and elderly subjects

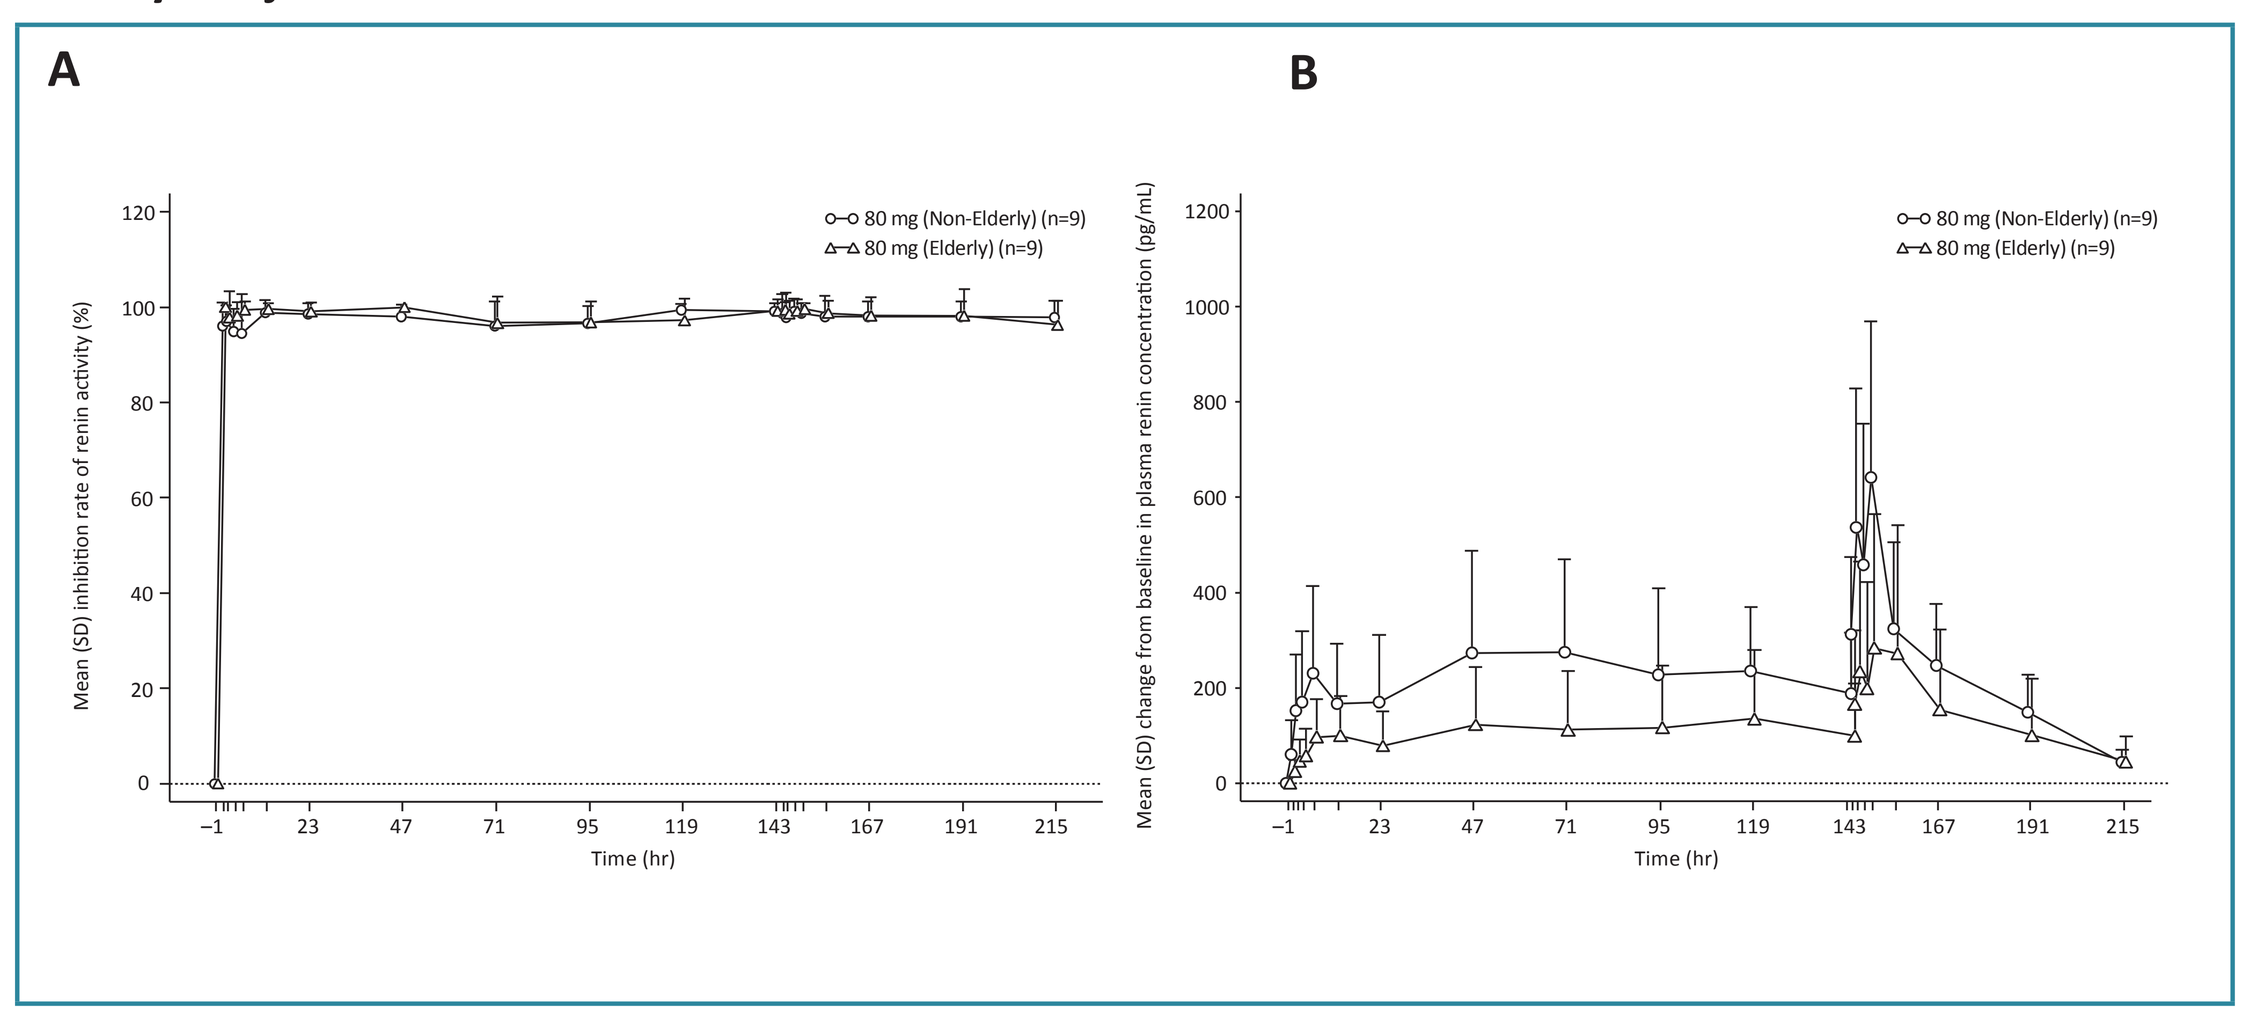
A
B
80 mg (Non-Elderly) (n=9)
80 mg (Elderly) (n=9)
Mean (SD) inhibition rate of renin activity (%)
Mean (SD) change from baseline in plasma renin concentration (pg/mL)
Time (hr)

PRA, plasma renin activity; PRC, plasma renin concentration; SD, standard deviation.

Safety

- There was no dose-dependent increase in the frequency of adverse events in non-elderly subjects (Table 5)
- The incidence of TEAEs was higher in elderly than in non-elderly subjects who received TAK-272 (33.3% vs 22.2%, respectively)
- TEAEs reported in ≥2 subjects were alanine aminotransferase increased (n=4), diarrhea (n=2), aspartate aminotransferase increased (n=2), and headache (n=2)
- All TEAEs were mild in intensity, with the exception of moderate urticaria in 1 non-elderly subject in the TAK-272 80-mg group. The outcomes of all events were “recovered/resolved”
- There were no serious TEAEs and no deaths during the study. No subject discontinued the study due to a TEAE
- No obvious changes were observed in the mean values of clinical laboratory tests, weight, or vital signs (except a decrease in mean blood pressure in the TAK-272 groups at Day 7 compared with baseline); no obvious changes were observed in ECG parameters and no clinically significant abnormalities were observed in ECG results at any timepoint

Table 5. Treatment-emergent adverse events (safety population)

	Non-elderly[a]			Elderly[b]	
Preferred term, n (%)	**Placebo (n = 6)**	**TAK-272 80 mg (n = 9)**	**TAK-272 160 mg (n = 3)**	**Placebo (n = 3)**	**TAK-272 80 mg (n = 9)**
Subjects with any TEAEs	1 (16.7)	2 (22.2)	2 (22.2)	1 (33.3)	3 (33.3)
Vision blurred	0	0	0	1 (33.3)	0
Diarrhea	0	0	0	1 (33.3)	1 (11.1)
Feeling hot	0	1 (11.1)	0	0	0
Alanine aminotransferase increased	1 (16.7)	0	2 (22.2)	0	1 (11.1)
Aspartate aminotransferase increased	1 (16.7)	0	0	0	1 (11.1)
Blood creatinine phosphokinase increased	1 (16.7)	0	0	0	0
Blood triglycerides increased	1 (16.7)	0	0	0	0
White blood cell count decreased	0	0	0	0	1 (11.1)
White blood cell count increased	0	1 (11.1)	0	0	0
Headache	0	0	0	1 (33.3)	1 (11.1)
Pruritus	0	1 (11.1)	0	0	0
Urticaria	0	1 (11.1)	0	0	0

[a]Subjects aged 20 to 45 years, inclusive.
[b]Subjects aged 65 to 85 years, inclusive.
TEAEs, treatment-emergent adverse events.

Conclusions

- This is the first study to evaluate the safety and PK/PD of multiple oral administrations of TAK-272 in human subjects
- During the period of multiple oral administrations of TAK-272 (80 or 160 mg):
 - The plasma concentration of TAK-272F reached steady state by Day 7
 - No PK accumulation was observed
 - PRA was rapidly inhibited at either dose; strong inhibition of PRA was observed for 7 days, and maintained for at least 71 hours after the last administration of TAK-272
- The AUC and C_{max} of TAK-272F were greater in elderly than in non-elderly subjects
- Multiple oral administrations of TAK-272 (80 or 160 mg) for 7 days were safe and well tolerated in non-elderly subjects; the 80-mg dose was also safe and well tolerated in elderly subjects
- Our results support future studies evaluating longer periods of TAK-272 administration, as well as studies evaluating the efficacy of TAK-272 in subjects with cardiovascular and/or renal disease

Disclosures

All authors are employees of Takeda Pharmaceutical Company, Ltd.

Acknowledgments

- The CPH-002 study was supported by Takeda
- Medical writing assistance was provided by BlueMomentum, an Ashfield business, part of UDG Healthcare PLC, and funded by Takeda

References

1. Imaeda Y, et al. *ACS Med Chem Lett*. 2016;7:933–938.
2. Matsuno K, et al. Safety, tolerability, pharmacokinetics (PK), and pharmacodynamics (PD) of single doses of TAK272, a novel renin inhibitor, in healthy male subjects [abstract]. 54th ERA-EDTA Congress; 2017 June 3–6; Madrid, Spain.

For further information contact Kumi Matsuno at kumi.matsuno@takeda.com.
Poster presented at the 54th ERA-EDTA Congress; Madrid, Spain; June 3–6, 2017.

54 ERA
428--SP
Diabetes - Clinical science
K Matsuno
DOI: 10.3252/pso.eu.54ERA.2017
ePosters supported by F. Hoffmann- La Roche Ltd.