HYPERPHOSPHATEMIA PROGRESSES CHRONIC KIDNEY DISEASE - MINERAL BONE DISEASE IN VASCULAR AND BONES: ESTABLISHMENT AND ANALYSIS OF NOVEL MOUSE MODEL.

Takashi Tani[1, 2], Hideo Orimo[2], Akira Shimizu[3], Shuichi Tsuruoka[1]

1. Department of Nephrology, Graduate School of Medicine, Nippon Medical School, Tokyo, Japan
2. Department of Metabolism and Nutrition, Graduate School of Medicine, Nippon Medical School, Tokyo, Japan
3. Department of Analytic Human Pathology, Graduate School of Medicine, Nippon Medical School, Tokyo, Japan

[Introduction] The prevalence of chronic kidney disease (CKD) is increasing due to global aging of society and an increased incidence of life-style related kidney disease. CKD - mineral bone disease (CKD-MBD), which includes ectopic calcification of arteries called medial arterial calcification (MAC) and renal osteodystrophy, is a serious complication and risk factor of CKD-related mortality. As mice, especially C57BL/6J strain, very commonly used in animal experiments, are quite resistant to ectopic calcification compared with other species, former CKD-MBD mouse models require surgical intervention or transgenic manipulation. (Topic: CKD-MBD)

[Objectives] To develop a novel, non-surgical, and non-transgenic mouse model of CKD using an adenine-fed procedure to investigate the clinical course of MBD.

[Methods] Eight-week-old C57/BL6J male mice were randomly allocated to the control, CKD-normal phosphorus (NP) and CKD- high phosphorus (HP) groups. Mice in the control groups, C6 (n=5) and C12 (n=5), were fed a standard chow containing 0.8% phosphorus (Pi) for 6 and 12 weeks, respectively. Mice in the CKD-NP groups, A6 (n=5) and A12 (n=5), were fed a chow containing 0.2% adenine and 0.8% Pi diet for 6 and 12 weeks, respectively. Mice in the CKD-HP mice were fed a chow containing 0.2% adenine and 0.8% Pi diet for 6 weeks, followed by a chow containing 0.2% adenine and 1.8% Pi for 2 weeks (A6P2 group, n=5), 4 weeks (A6P4 group, n=5) and 6 weeks (A6P6 group, n=5). At sacrifice, plasma and urine were collected to measure serum and urine calcium, phosphorus, creatinine, urea levels, intact parathyroid hormone, fibroblast growth factor 23 and 24-h creatinine clearance. Aortic tissue was harvested, and evaluated by histological and mRNA analysis. Aorta and femur bone were scanned by computed tomography (CT) imaging for quantitative evaluation.

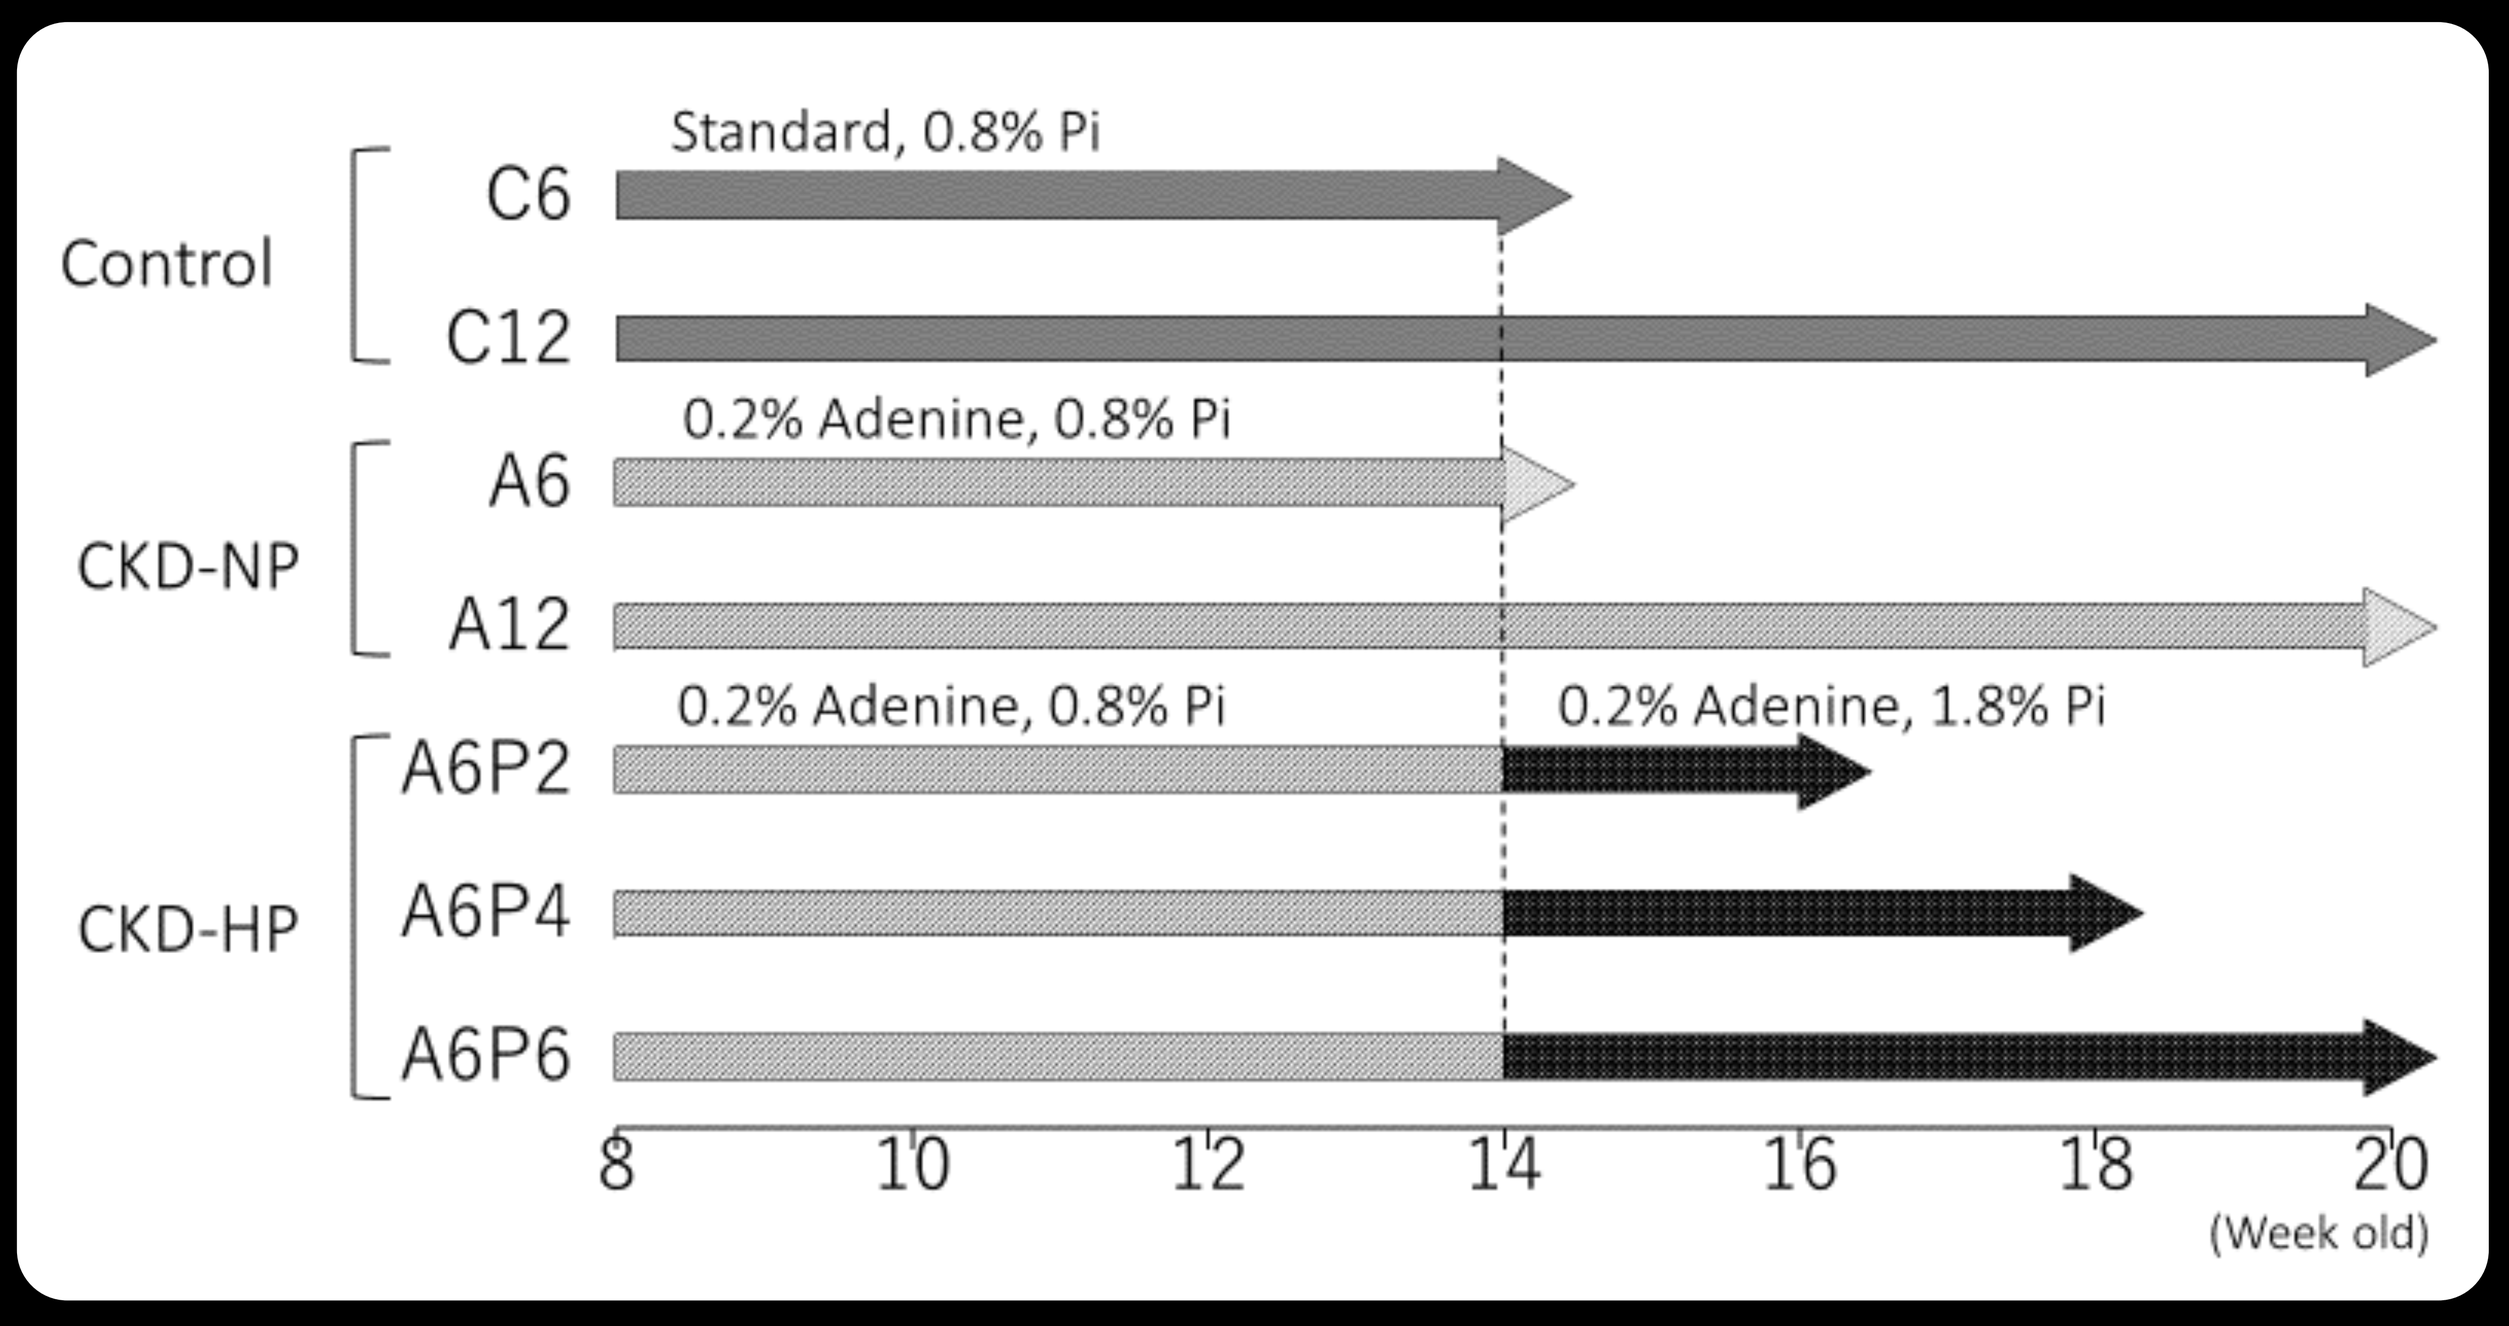

Control
C6
Standard, 0.8% Pi
C12
CKD-NP
A6
0.2% Adenine, 0.8% Pi
A12
CKD-HP
A6P2
0.2% Adenine, 0.8% Pi
0.2% Adenine, 1.8% Pi
A6P4
A6P6
8
10
12
14
16
18
20
(Week old)

[Results] Blood levels of urea nitrogen and serum creatinine were elevated in both CKD-NP and CKD-HP group, while serum phosphorus and intact parathyroid hormone were significantly increased in the CKD-HP than CKD-NP mice (**Table 1.**). There was clear evidence (positive Von-Kossa and Alizarin Red staining) of MAC exclusively in the media, and not the adventitia, in CKD-HP mice (**Fig. 1a-d**). Computed tomography (CT) imaging of CKD-HP mice confirmed MAC mainly in the abdominal aorta, up to the aortic bifurcation (**Fig. 1e,f; arrows**). Also, the volume of aortic calcification increased with longer exposure to the high phosphorus feed evaluated with CT and histological imaging (**Table 1.**). MAC formation in CKD-HP mice was associated with upregulation in runt-related transcription factor 2 (Runx2), tissue non-specific alkaline phosphatase (TNAP) and osteopontin (OPN), indicative of osteoblastic trans-differentiation of vascular smooth muscle cells (**Fig. 1i-iii**). CT imaging and histology revealed that the cortex of the femur was thinned in the CKD group (**Fig. 2d-i**), particularly in CKD-HP mice (**Fig. 2g-i**), compared to the thick and homogeneous appearance of the wild-type murine cortical bone (**Fig. 2a-c**). The morphometric parameters evaluated from CT imaging of the distal femoral metaphysis showed significant depletion in bone mineral density/volume in all CKD group, while the depletion in the cortical bone was remarkable in CKD-HP mice than CKD-NP and control (**Fig. 2i,ii**).

[Conclusion] Our results support a strong influence of hyperphosphatemia on MAC formation and renal osteodystrophy, which is consistent with results from clinical studies describing a close association between serum phosphorus levels, vascular calcification formation and mortality among patients with CKD. Our novel CKD-MBD model is enhanced by its recapitulation of CKD-related complications of arterial calcification and renal osteodystrophy in patients with end-stage renal disease in practice, and we expect this model to be of value advancing research in the field of CKD.

Table1. Serum, urine biochemistries and quantification of MAC in CKD and control mice.

	Control		CKD				
			CKD-NP		CKD-HP		
	C6	C12	A6	A12	A6P2	A6P4	A6P6
BUN (mg/dl)	32.0±6.3	32.8±4.3	125.1±9.5*	159.9±11.2$^{*\#}$	85.2±4.4$^{*\#\delta}$	75.6±9.8$^{*\#\delta}$	70.3±7.0$^{*\#\delta}$
Creatinine (mg/dl)	0.12±0.03	0.10±0.02	0.66±0.04*	0.91±0.11$^{*\#}$	0.47±0.06$^{*\#\delta}$	0.41±0.05$^{*\#\delta}$	0.42±0.06$^{*\#\delta}$
Calcium (mg/dl)	8.08±0.59	8.21±0.18	8.52±0.79	9.14±0.80	8.66±0.91	8.13±0.80	8.19±1.18
Phosphorous (mg/dl)	8.01±2.70	7.43±1.67	9.47±2.14	12.3±2.40*	20.9±1.90$^{*\#\delta}$	18.7±4.55$^{*\#\delta}$	18.9±3.44$^{*\#\delta}$
intact PTH (pg/ml)	112±53	172±60	1482±498*	2854±984*	4503±689$^{*\#}$	4214±820$^{*\#}$	4150±813$^{*\#}$
FGF-23 (pg/ml)	104±18	115±12	2554±892*	5415±694*	4508±1102*	4040±951*	5666±3704*
Calcified volume by qCT analysis	Aorta (mm^3)	0.28±0.07[a]	0.27±0.02[a]	0.32±0.13[a]	0.67±0.78$^{*\#}$	2.38±3.00$^{*\#}$	2.90±3.98$^{*\#}$
	Heart (mm^3)	0.35±0.16[a]	0.14±0.06[a]	0.21±0.07[a]	0.42±0.24$^{*\#}$	3.25±2.78$^{*\#}$	2.81±2.47$^{*\#}$
OCPC assay [Ca^{2+}] [b]		0.01±0.00	0.01±0.04	0.02±0.03	0.05±0.01	1.4±0.1$^{*\#\delta}$	5.4±0.2$^{*\#\delta}$
Percentage calcified surface in aorta (%)[c]		N.D	N.D	N.D	5.0±10.4	11.8±14.8	15.7±12.8

MAC, medial arterial calcification; BUN, blood urea nitrogen; 24 h CCr, 24 hour creatinine clearance; FEphos, fractional excretion of phosphorus; ALP, alkaline phosphatase; PTH, parathyroid hormone; FGF-23, fibroblast growth factor 23; qCT, quantitative computed tomography; OCPC, o-cresolphthalein complexone; Ca, calcium; N.D: not detected. a, calcified volume in control and CKD-NP groups are considered to be false positive by mechanical artifacts; b, umol/mg dry weight; c, % Calcified surface (%).

Figure 1 a-e. Histological and CT images of the aorta in CKD-HP mice.

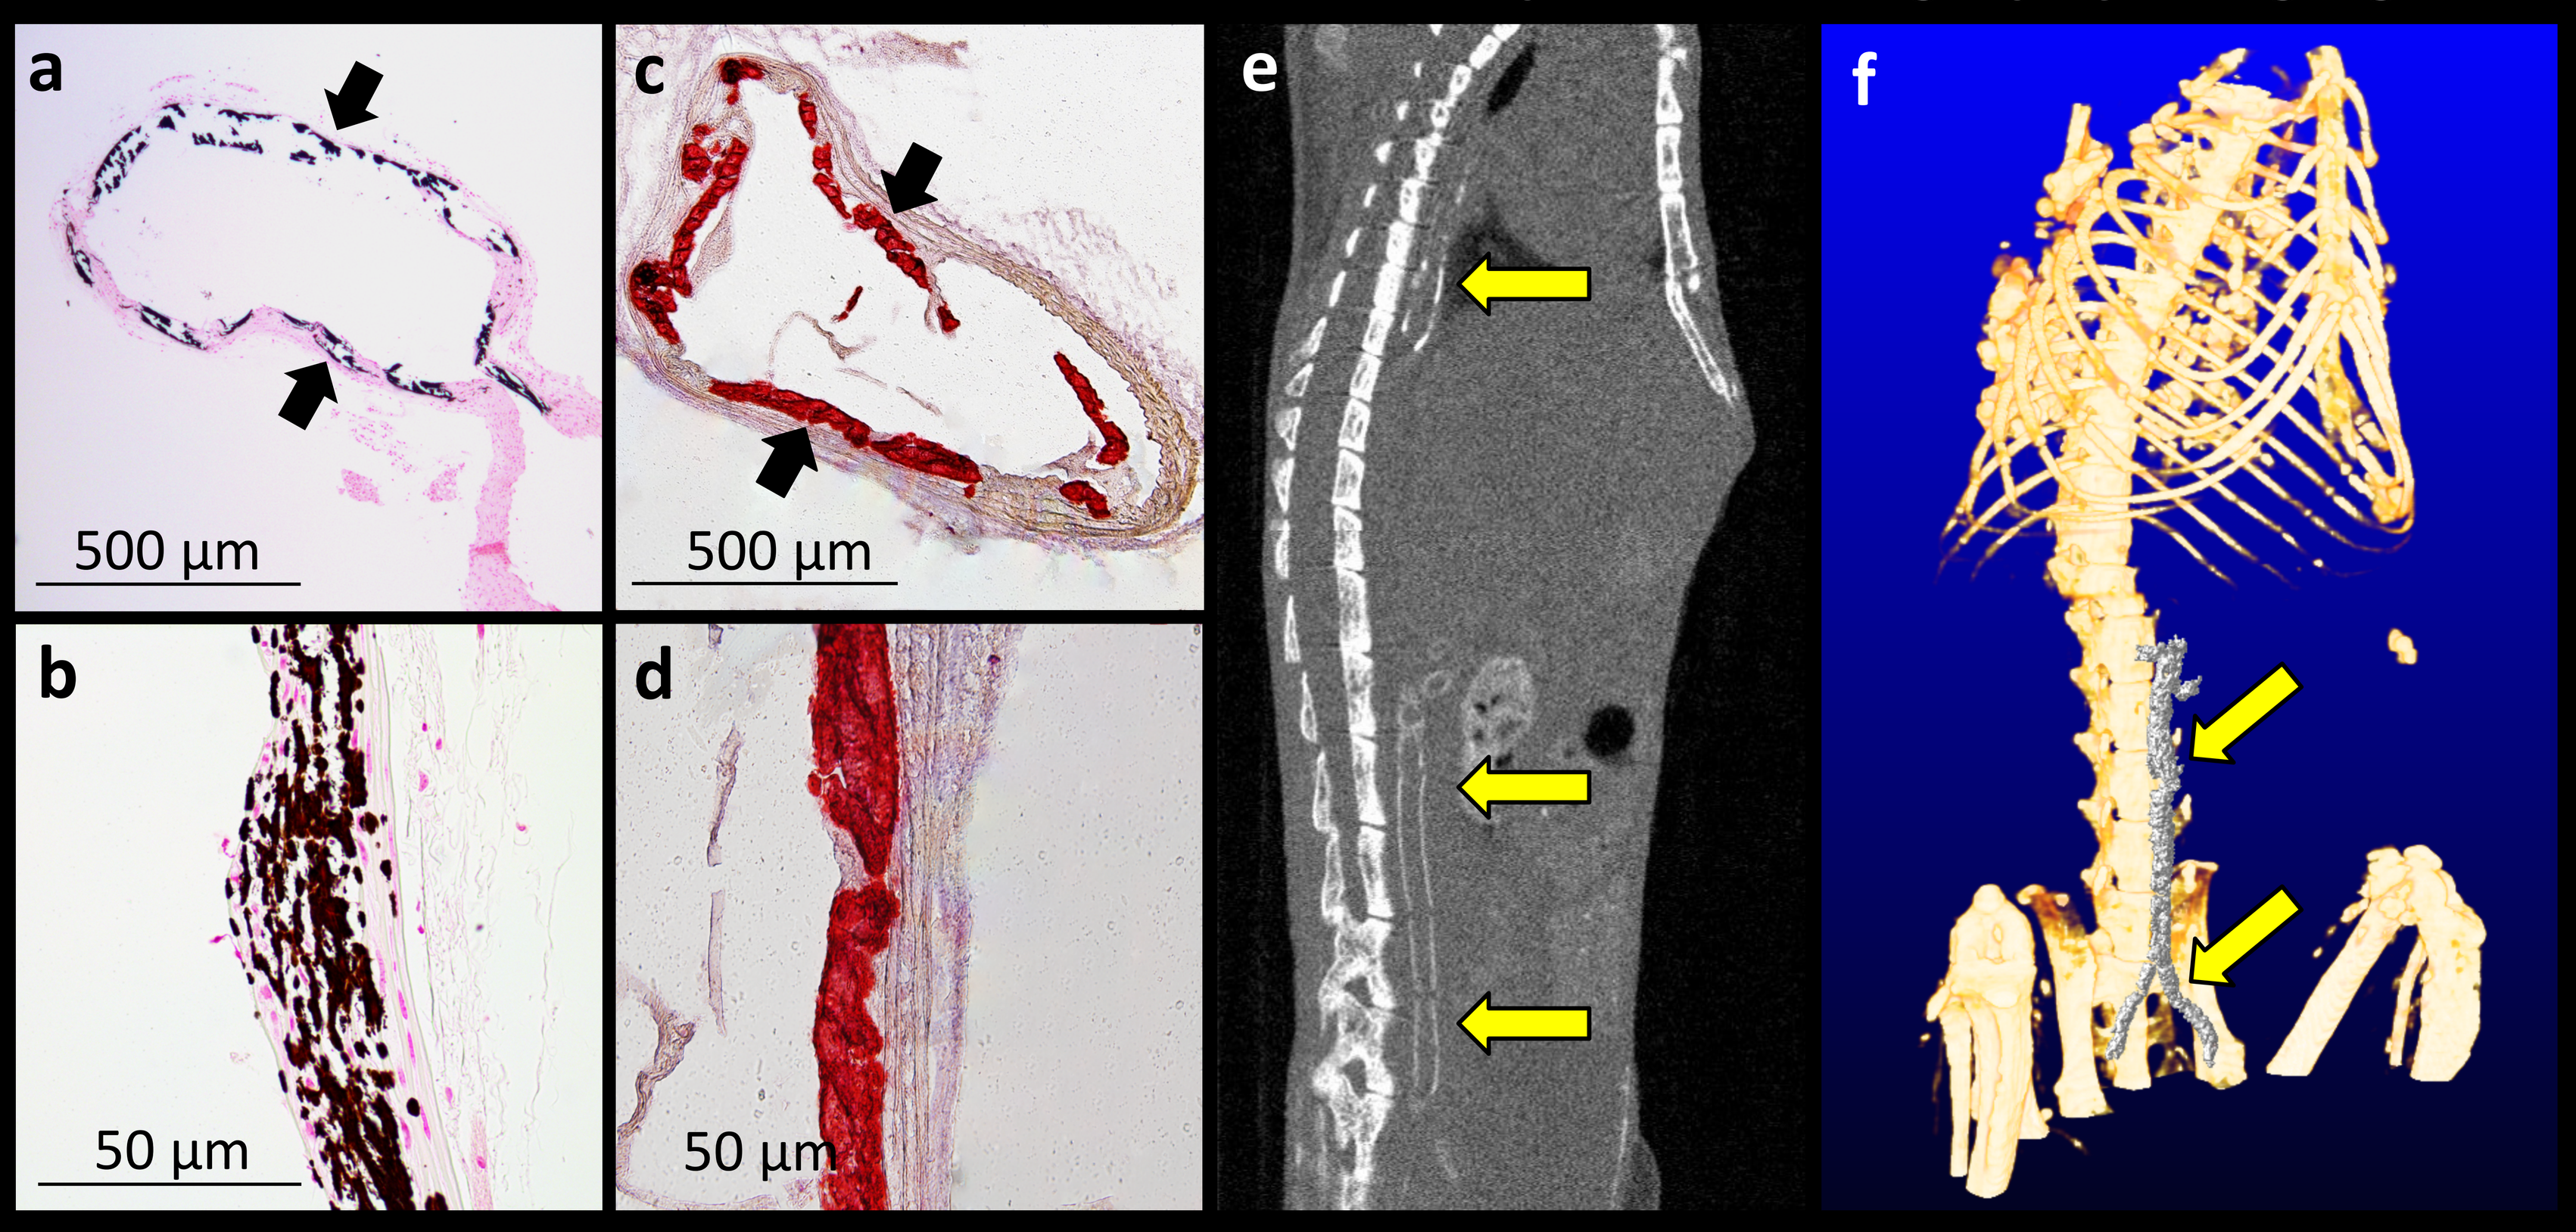

Von-Kossa
Alizarin Red
Computed Tomography imaging
a
500 µm
c
500 µm
e
f
b
50 µm
d
50 µm

Figure 1 (i)-(iii). Quantitative real-time RT-PCR analysis of aortic tissue.

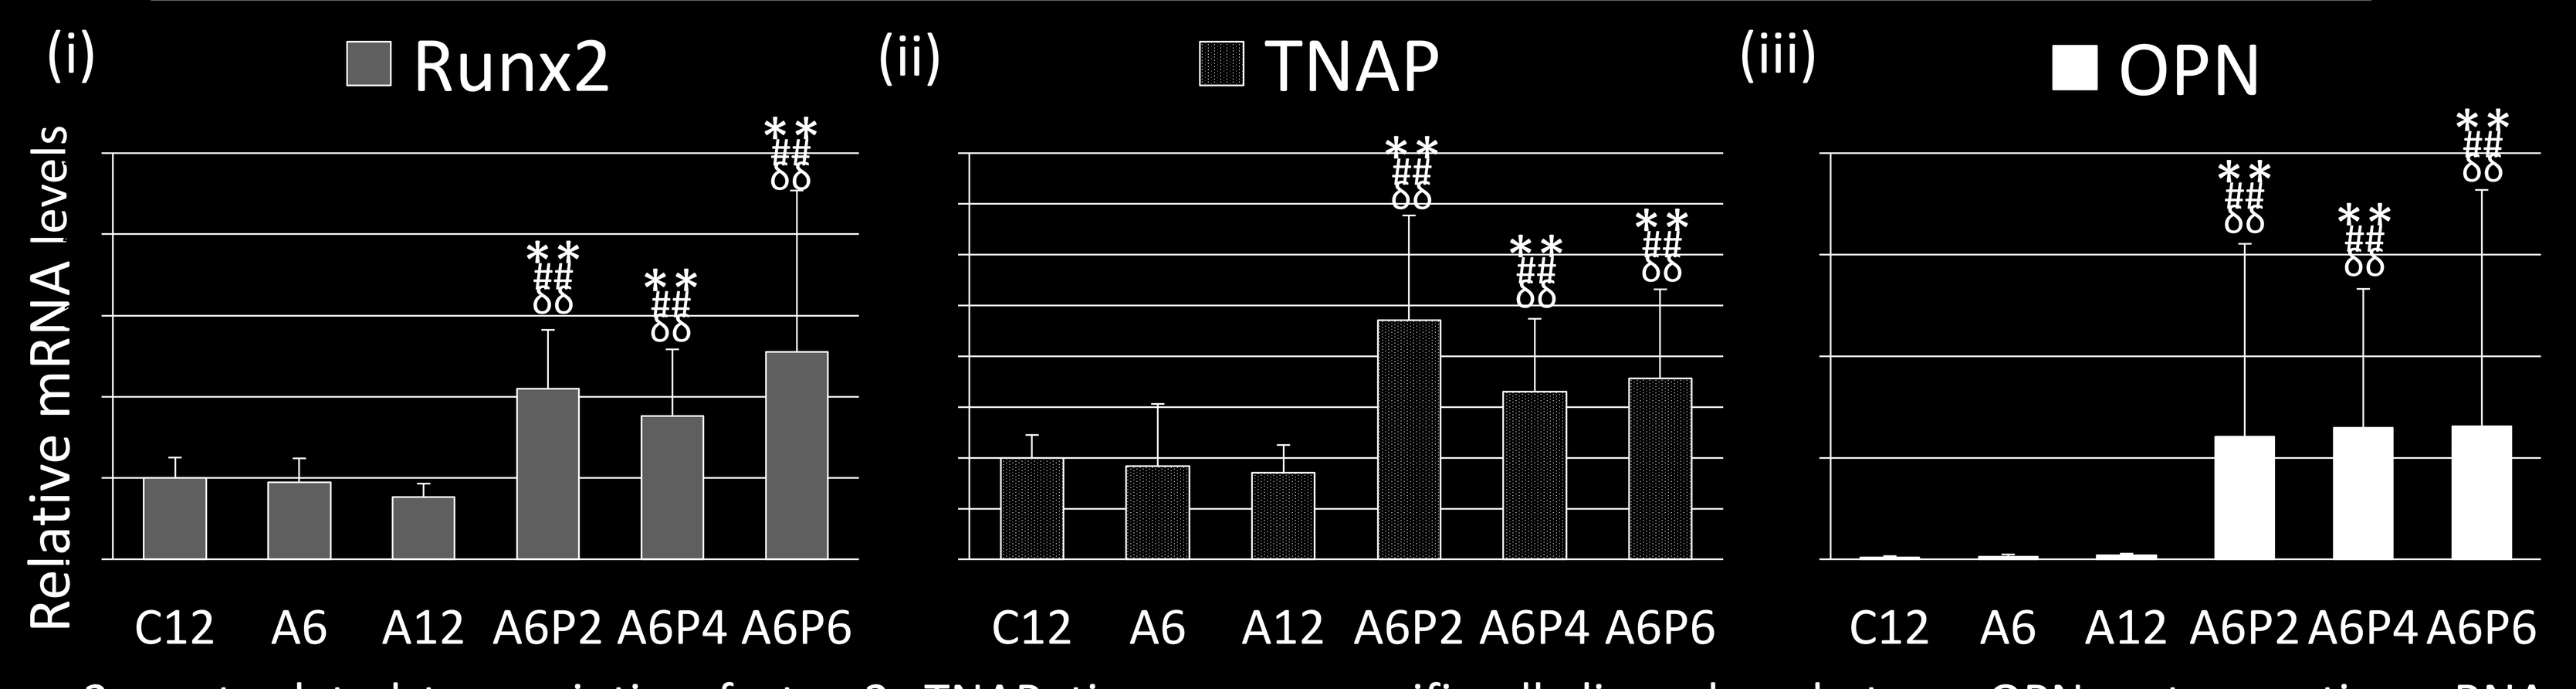

(i)
Runx2
(ii)
TNAP
(iii)
OPN
Relative mRNA levels
C12 A6 A12 A6P2 A6P4 A6P6

Runx2, runt-related transcription factor 2; TNAP, tissue non-specific alkaline phosphatase; OPN, osteopontin. mRNA were determined by quantitative real-time PCR in triplicate and normalized to 18S rRNA levels.

Figure 2 a-i. Histological and CT images of the distal thighbone in CKD-HP mice.

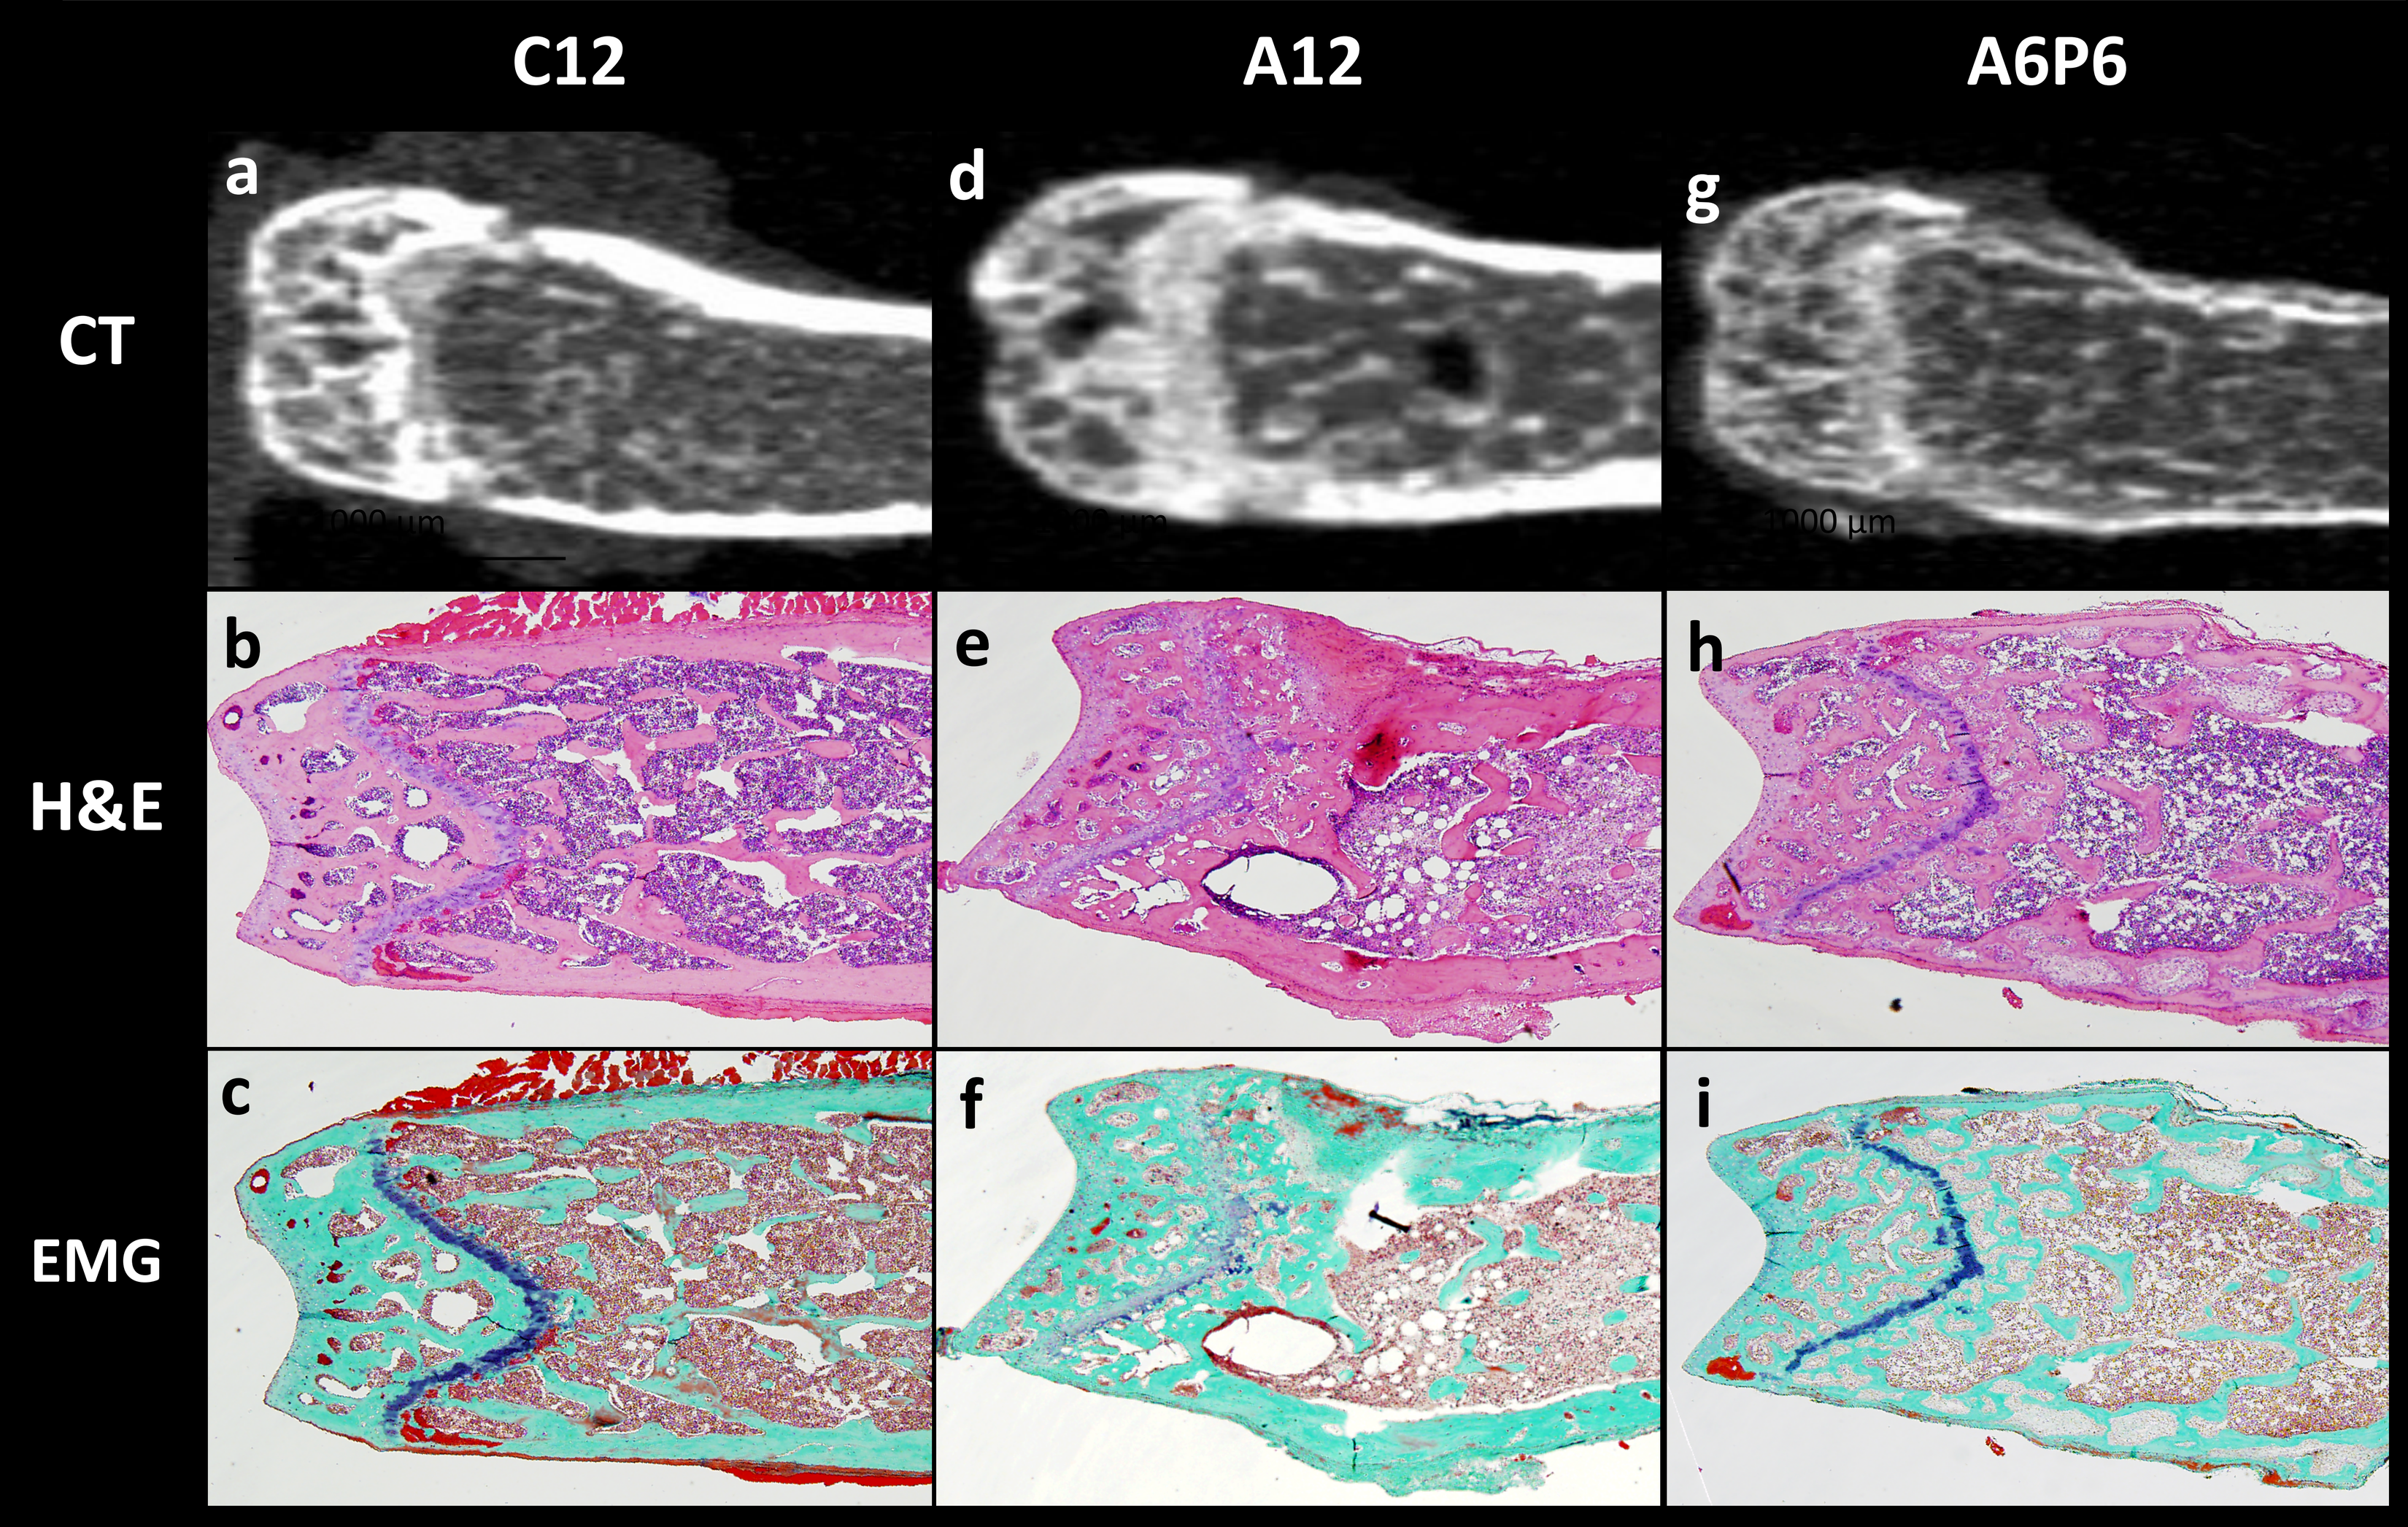

C12
A12
A6P6
CT
H&E
EMG
a
d
g
b
e
h
c
f
i

Figure 2 (i)-(iii). Morphometric parameters of distal femoral metaphysis.

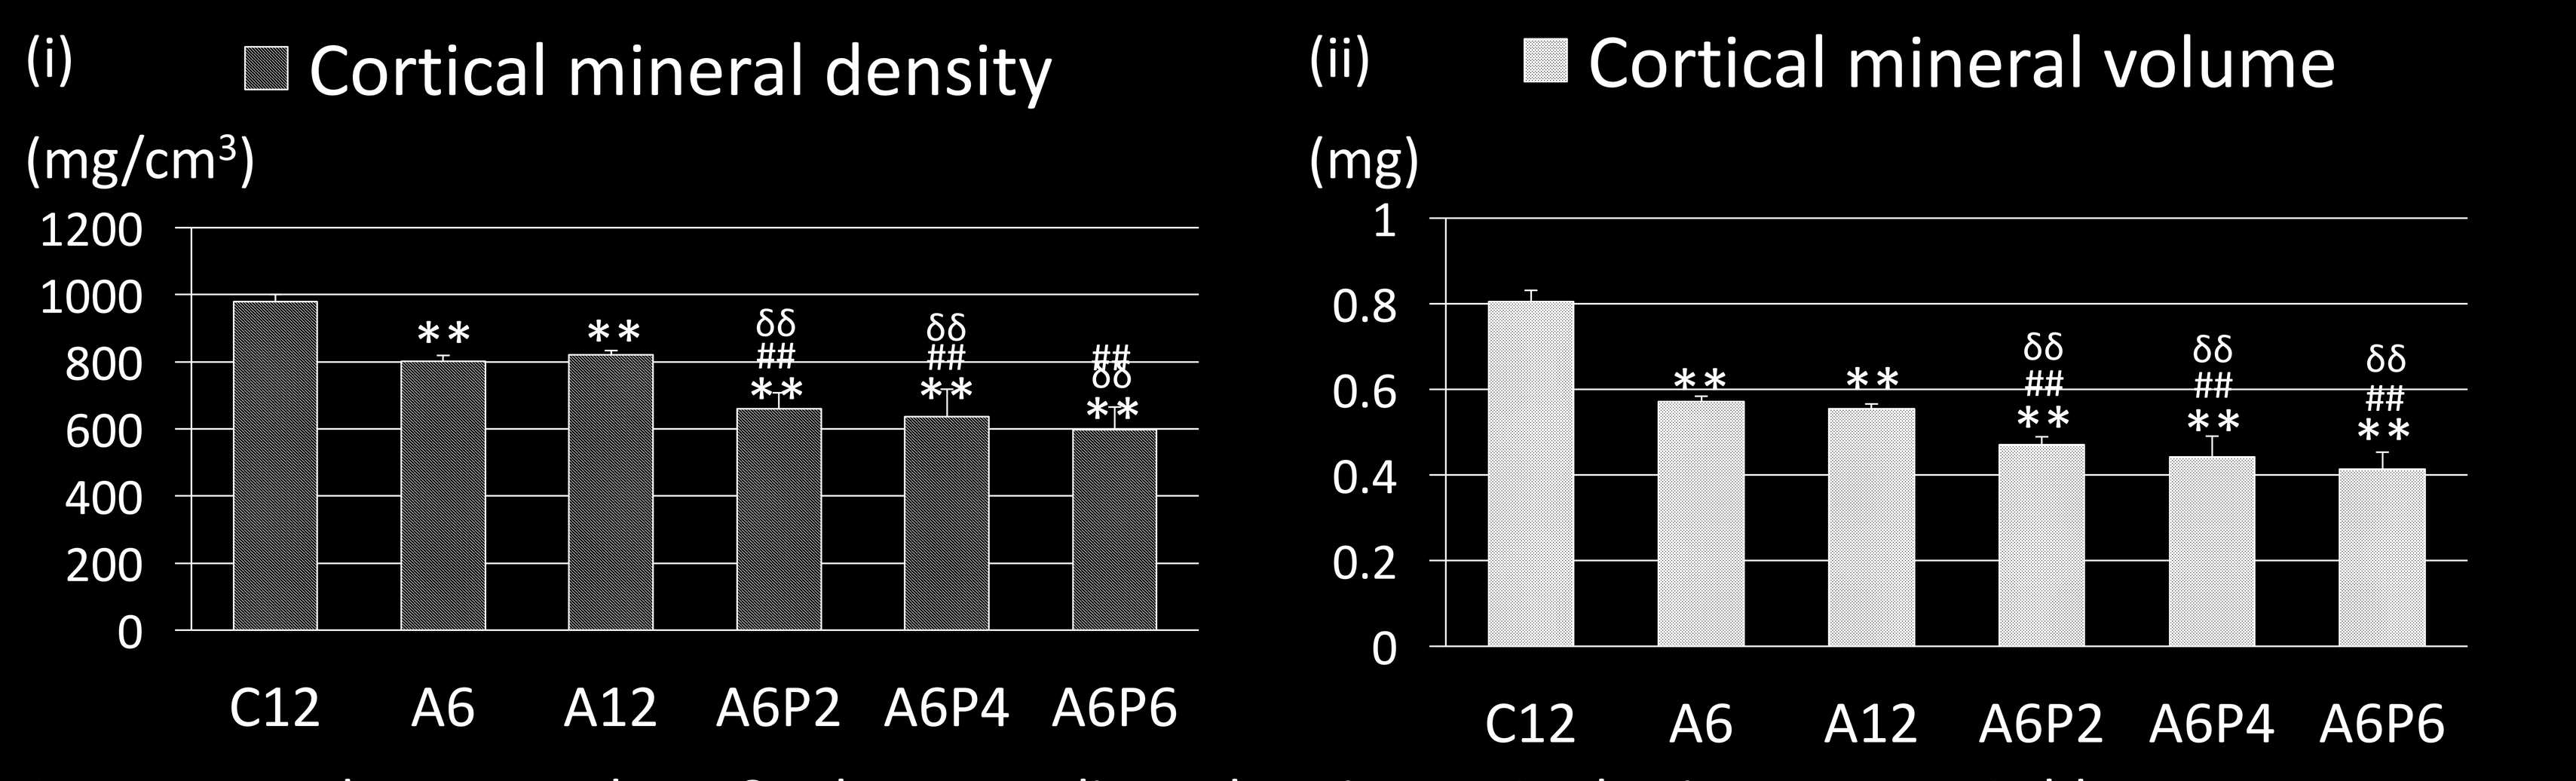

(i)
Cortical mineral density
(mg/cm^3)
1200
1000
800
600
400
200
0
C12 A6 A12 A6P2 A6P4 A6P6
(ii)
Cortical mineral volume
(mg)
1
0.8
0.6
0.4
0.2
0
C12 A6 A12 A6P2 A6P4 A6P6

CT, computed tomography; H&E, hematoxylin and eosin; EMG, elastica Masson-Goldner.

All the data above are represented as the means ± SEM (n = 5). $^{*}P < 0.05$, $^{**}P < 0.01$ vs C6 and/or C12 by non-repeated measures ANOVA. $^{\#}P < 0.01$, $^{\#\#}P < 0.01$ vs A6 by non-repeated measures ANOVA. $^{\delta}P < 0.05$, $^{\delta\delta}P < 0.01$ vs A12 by non-repeated measures ANOVA.

54 ERA
359--SP
CKD - MBD
Takashi Tani

DOI: 10.3252/pso.eu.54ERA.2017

ePosters supported by F. Hoffmann- La Roche Ltd.