Incidence and determinants of hyperkalemia in a large North European healthcare system

Erik Nilsson[1, 2], Alessandro Gasparini[1,3], Johan Ärnlöv[4], Hairong Xu[5], Karin M Henriksson[4], Josef Coresh[6], Morgan E Grams[6], Juan J Carrero[1]

[1]Divisions of Renal Medicine and Baxter Novum, Department of Clinical Science, Intervention and Technology, Karolinska Institutet, Stockholm, Sweden; [2]School of Medical Sciences, Örebro University, Örebro, Sweden; [3]Department of Health Sciences, University of Leicester, United Kingdom; [4]Department of Medical Sciences, Uppsala University Hospital, Uppsala, Sweden; [5]AstraZeneca, Gaithersburg, MD, USA; [6]Department of Epidemiology, Johns Hopkins Bloomberg School of Public Health, Baltimore, Maryland, USA

Introduction and aims

The burden of hyperkalemia in clinical settings is insufficiently characterized. We studied hyperkalemia incidence and associated risk factors in a large Swedish healthcare system.

Methods

Observational study from the Stockholm CREAtinine Measurements (SCREAM) project, including adult healthcare users (n=364 955) in the region of Stockholm, Sweden, undergoing potassium testing during 2009 and with a recent serum creatinine test to estimate glomerular filtration rate (eGFR). Over a 3-year observation period, the incidence proportion and rates of hyperkalemia were estimated overall, by selected comorbid conditions and by medication use. Hyperkalemia was defined as serum potassium >5 mmol/L, and moderate/severe hyperkalemia as serum potassium >5.5 mmol/L. Kidney function was defined by the following eGFR strata: G1-2= eGFR≥60; G3= eGFR 60-30 and G4+= eGFR<30 ml/min/1.73m2.

Results

Over the 3-year period, hyperkalemia occurred in 25 461 (7%) individuals. Most cases (65%) had only one event during follow-up. Moderate/severe hyperkalemia occurred in 9059 (2.5%) individuals. Again, most cases (72%) occurred only once. The incidence proportion of hyperkalemia was higher in the presence of specific comorbidities and among users of renin angiotensin-aldosterone system inhibitors (**Figure 1**). For instance, 55% of patients with eGFR G4+ and 24% of patients with heart failure developed hyperkalemia within 3 years. The incidence rate of hyperkalemia was 49.9/1000 person years, with considerably higher rates observed in those with diabetes, lower eGFR (**Table 1**), myocardial infarction, heart failure or use of renin-angiotensin-aldosterone system inhibitors. In multivariable logistic regression analysis, female sex and baseline use of loop/thiazide diuretics associated with lower hyperkalemia risk while older age categories, lower eGFR strata, diabetes mellitus, heart failure and baseline use of angiotensin converting enzyme inhibitors, angiotensin receptor inhibitors and mineralocorticoid receptor antagonists associated with higher risk (**Table 2**).

Conclusions

Hyperkalemia is common in healthcare. Optimal RAASi use and careful potassium monitoring in the presence of certain comorbidities, especially lower eGFR, is advocated.

Acknowledgements

This study was supported by an institutional grant from AstraZeneca to Karolinska Institutet. We acknowledge other grant support from the Swedish Heart and Lung Foundation, the Stockholm County Council, Vifor Fresenius Medical Care Renal Pharma, Martin Rind's and Westman's Foundations.

Incidence rates of hyperkalemia by eGFR

Category	Crude IR (95% CI)	Adjusted IR (95% CI)
G1-2	22.6 (22.3-23)	10.8 (10.5-11.1)
G3	156 (154-159)	30.5 (29.1-31.9)
G4+	1070 (1050-1090)	133 (123-144)

Table 1: Analysis was based on patients with at least one potassium measurement. Per 1,000 person years. Adjusted IRs are obtained from a zero-inflated negative binomial model, and are adjusted for age category, gender, renal function, history of heart failure, history of myocardial infarction, hypertension status, diabetes mellitus, use of RAASi, and number of K+ tests per year as a categorical variable. Adjusted IRs are presented fixing the value of the adjustment factors to their mean value. Abbreviations: eGFR, estimated glomerular filtration rate.

Cumulative risk of hyperkalemia

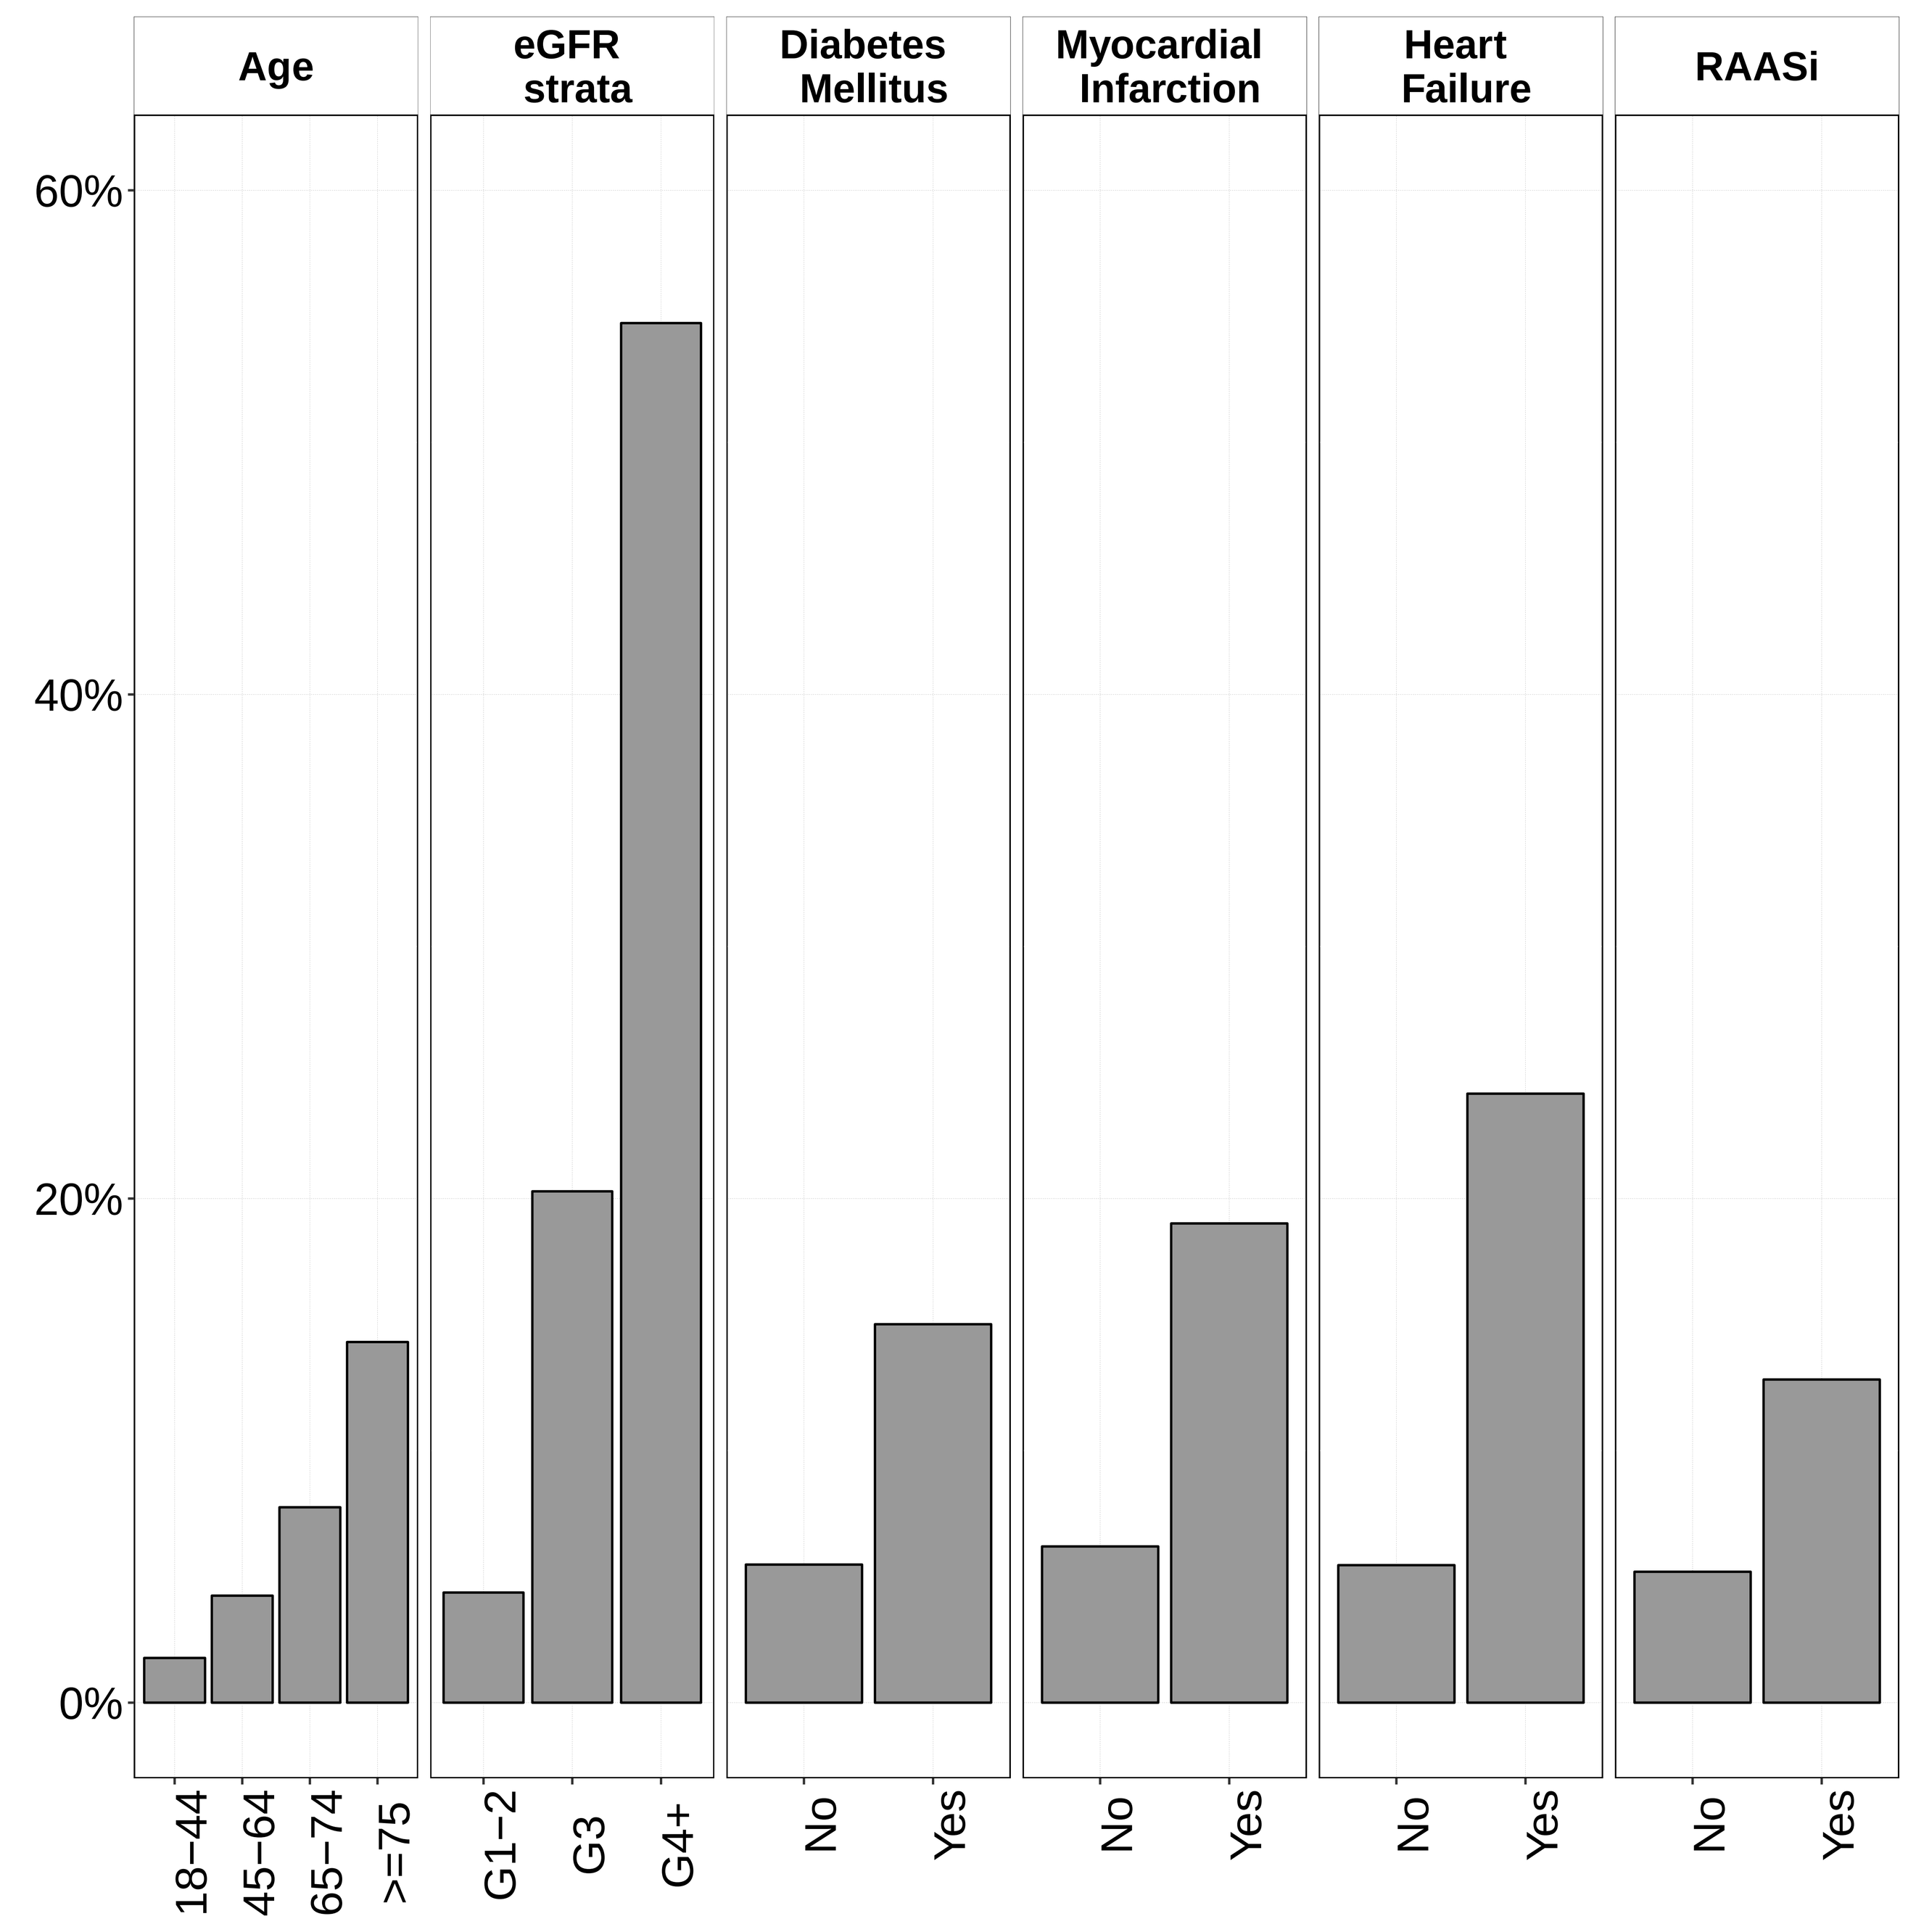
Age
eGFR strata
Diabetes Mellitus
Myocardial Infarction
Heart Failure
RAASi
60%
40%
20%
0%
18–44
45–64
65–74
>=75
G1–2
G3
G4+
No
Yes
No
Yes
No
Yes
No
Yes

Figure 1: Three-year incidence proportion of hyperkalemia (K > 5.0 mmol/L). Abbreviations: RAASI, Renin-Angiotensin-Aldosterone System Inhibitor.

Risk factors for hyperkalemia

Factor	Odds Ratio (95% CI)
Age: 18-44	Ref.
Age: 45-64	1.52 (1.42-1.63)
Age: 65-74	1.82 (1.69-1.95)
Age: >=75	1.87 (1.74-2.01)
Gender: female	0.79 (0.77-0.81)
eGFR: G1-2	Ref.
eGFR: G3	2.14 (2.07-2.22)
eGFR: G4+	5.6 (5.24-5.98)
Diabetes mellitus	1.73 (1.67-1.79)
Hypertension	1.05 (1-1.1)
Heart failure	1.14 (1.09-1.19)
Myocardial infarction	1.03 (0.98-1.08)
Peripheral vascular disease	1.22 (1.15-1.28)
Cerebrovascular disease	0.97 (0.93-1.01)
ACEi use	1.51 (1.46-1.57)
ARBs use	1.21 (1.17-1.26)
MRAs use	1.66 (1.41-1.95)
Beta-blockers use	1.06 (1.03-1.1)
NSAIDs use	1 (0.96-1.04)
Loop/Thiazide diuretics use	0.94 (0.9-0.97)
K-sparing diuretics	1.08 (0.92-1.26)
Other blood pressure medications use	0.88 (0.85-0.91)

Table 2: Risk factors for hyperkalemia (>5.0 mmol/L). Adjusted odds ratios with 95% confidence intervals from logistic regression on hyperkalemia. Adjustment was for age category, female, eGFR category, diabetes mellitus, hypertension, chronic heart failure, myocardial infarction, peripheral vascular disease, cerebrovascular disease, ace inhibitor, angiotensin receptor inhibitor, mineralocorticoid receptor blocker, beta blocker, non-steroidal antinflammatory drug use, loop- or thiazide diuretic, potassium sparing diuretic, other blood pressure medication, potassium samplings per year. Abbreviations: eGFR, estimated glomerular filtration rate; ACEi, angiotensin converting enzyme inhibitors; ARB, angiotensin receptor inhibitors; MRA, mineralocorticoid receptor antagonists; NSAID, non-steroidal antiinflammatory drugs.

potassium (n.) Metallic element, 1807, coined by English chemist Sir Humphrey Davy (1778-1829) from Modern Latin *potassa*, Latinized form of *potash* (q.v.). Davy first isolated it from potash. Symbol K is from Latin *kalium* "potash," from Arabic *al-qaliy* "the ashes, burnt ashes" [Source: etymonline.com]

Contact: erik.nilsson2@regionorebrolan.se

54 ERA
313--SP
CDK - Clinical epidemiology I
Erik Nilsson
DOI: 10.3252/pso.eu.54ERA.2017
ePosters supported by F. Hoffmann- La Roche Ltd.