Higher risk of renal disease in chronic hepatitis C patients, and treatment improved survival for hepatitis C patients on hemodialysis – A nationwide population-based register study from 2001 to 2013

Annette Bruchfeld[1], Charlotta Millbourn[2], Karin Lindahl[2], Jan Kövamees[3], Robert Schvarcz[2], Katharina Büsch[3,4], and Jonas Söderholm[3,5]

[1]Department of Renal medicine, CLINTEC, Karolinska Institutet, Stockholm Sweden, [2]Department of Medicine, Division of Infectious Diseases, Karolinska Institutet at Karolinska University Hospital, Stockholm, Sweden, [3]AbbVie AB, Stockholm, Sweden, [4]Department of Medicine, Karolinska Institutet, Stockholm, Sweden, [5]Department of Laboratory Medicine, Division of Clinical Microbiology, Karolinska Institutet at Karolinska University Hospital Huddinge, Stockholm, Sweden

Presented at the ERA-EDTA 2017, June 3–6, 2017, Madrid, Spain

INTRODUCTION

Approximately 75%-85% of Hepatitis C Virus (HCV) infected patients develop a chronic HCV infection (Chen and Morgan, 2006). Besides the negative impact by the virus on the liver, leading to liver cirrhosis in 40 % of cases after 30 years (Thein *et al.*, 2008), HCV also affect other organs with a 70% higher risk of a chronic kidney disease (CKD) stage 3 (defined as eGFR <60mL/min/1.73m^2) in HCV seropositive patients compared with seronegative patients (Fabrizi *et al.*, 2014).

The seroprevalence of HCV in dialysis patients has been shown to be between 3-20% in Western Europe and the United States (Martin and Fabrizi, 2008). HCV treatment of hemodialysis (HD) patients using interferon-α (IFN) with or without ribavirin (RBV) has been cumbersome due to adverse events.

In a large international observational study, it was shown that only 1.0% of HCV patients on HD had received anti-viral treatment (Goodkin *et al.,* 2013). The standard of care for treating HCV has been changing rapidly over the last few years with several IFN-free options approved or in phase 3 studies (Gane et al., 2016; Pockros *et al.*, 2016; Roth *et al.*, 2015;).

OBJECTIVE

- Describe the CKD population within the Swedish patients with HCV.
- Describe the Swedish HCV patients undergoing HD.
- Investigate the impact of IFN-based treatment on survival in HCV HD patients.

METHODS

DATA SOURCES

The Swedish nationwide registers were used to identify patients with chronic HCV as well as to evaluate the occurrence of other diagnosis or use of medical procedures . Register sources included the National Patient Register, the Cancer Register and the Prescribed Drug Register, all kept by the Swedish National Board of Health and Welfare (**Table 1**). Data on place of residence, vital and emigration status were retrieved from the Register of the Total Population held by Statistic Sweden (up to December 31, 2013). This register covers the entire Swedish population and includes information on age, sex, and place of residence as well as dates of birth, death, and emigration status. The Swedish personal identity number (social security number) was used to link individuals between registers. Up to five general population comparators were matched by age, sex, and county of residence to each patient at time of diagnosis / identification (n=202,694). The two cohorts were recently described elsewhere (Büsch *et al*., 2016:1). The study was approved by the Regional Ethics Committee, Karolinska Institutet, Stockholm, Sweden.

Table 1. Description of national Swedish registers

Register	Description
National Patient Register	contains all in-patient and non-primary outpatient care visits such as treatment visits to an infectious disease specialist or gastroenterologist, but no primary care data. Available register data from: In-patient care 1987–2013; Day surgery 1997–2000; and Non-primary outpatient care 2001–2013 (including day surgery). It includes information on main and contributory diagnoses based on the International Classification of Diseases (ICD-9 1987–1996; ICD-10 1997-2013).
Cancer Registry	covers medical data such as the site of tumor, histological type, basis and date of diagnosis. The register covers the whole population and it is compulsory for every health care provider to report newly detected cancer cases to the registry thus it is generally considered to be of good quality as approximately 99% of the cases are morphologically also verified.
Prescribed Drug Register	registers all dispensed prescribed drug use in ambulatory care using Anatomical Therapeutic Chemical (ATC) codes. This register retains information on dates, drugs and costs for all pharmacy dispensations of prescriptions in Sweden (2005–2013). The coverage is complete for prescriptions in ambulatory care, while in-hospital use of drugs is captured to a lesser extent.

ABBREVIATIONS

ATC - Anatomical Therapeutic Chemical; **CI** - Confidence Interval; **HCV** - Chronic Hepatitis C; **CKD** - Chronic Kidney Disease; **DAA** - Direct Acting Antiviral; **DSV** – Dasabuvir; **EBR/GZR** - Elbasvir/Grazoprevir; **HCV** - Hepatitis C Virus; **HD** - Hemodialysis; **ICD** - International Classification of Diseases; **IFN** – Interferon; **OBV/PTV/r** - Ombitasvir/Paritaprevir/Ritonavir; **RBV** – Ribavirin; **SIR** - Standardized Incidence Ratio; **SMR** - Standardized Mortality Ratio; **SUR** - Standardized Utilization Ratio; **SVR** - Sustained Virological Response

METHODS (CONTINUED)

SETTING

In Sweden, universal access to health care is provided to the population through a tax funded system. HCV patients are typically cared for by specialists in infectious diseases or gastroenterology in hospital based outpatient clinics or inpatient facilities. They are not cared for by general practitioners in primary care .

IDENTIFICATION OF PATIENTS AND MATCHED GENERAL POPULATION CONTROLS

Diagnosis were identified in the National Patient Register, the Cancer registry, and treatments in the Prescribed Drug Registry using:

<u>ICD-10 codes</u>
Chronic Hepatitis C (B18.2), Acute Kidney Failure (N17), Chronic Kidney Disease (N18), Unspecific Kidney Failure (N19), Kidney Transplantation (Z94.0), Cryoglobulinemia (D89.1), Diabetes (E10-E14)
<u>ICD-7 codes</u>
Kidney Cancer (180.0/180.9)
<u>Procedure codes</u>
Hemodialysis (DR016)
<u>ATC codes for dispensed prescribed drugs</u>
Diabetes (A10), HCV treatment (L03AB11, L03AB60, L03AB61, J05AB04, J05AE11, J05AE12)

OBSERVATION TIME

Since the non-primary outpatient care data started being included in the National Patient Register in 2001 this year was the starting point in the present study. The observation time started for the HCV cohort (n=42,522) at the time of first physician visit with an accompanying HCV (B18.2) ICD-code between 2001 and 2013. These index dates were also used for each comparator (n=202,694). The observation time ended at the time of death, emigration, or the 31st of December 2013, whichever came first.

ASSESSMENTS

The risk for diseases, procedures, or mortality were expressed as standardized ratios, where the number of observed events was divided by the number of expected events in the HCV cohort based on the events per person-years in the comparator cohort. Disease incidences were shown as Standardized Incidence Ratios (SIR), the procedure events were shown as Standardized Utilization Ratio (SUR), and the mortalities were shown as Standardized Mortality Ratio (SMR). All standardized ratios were shown in combination with 95% Confidence Intervals (CI).

HEMODIALYSIS

The procedure code for HD was not used before 2005, and the Prescribed Drug Registry was only available from 2005. Therefore neither HCV treatment nor HD between 2001 and 2004 could not be assessed in the present study. The annual number of patients on in-hospital HD in Sweden (2,285 to 2,881 between 2010 and 2013) was retrieved from the annual report by the Swedish Kidney Registry (SNR, 2014).

STATISTICAL METHODS

Data handling was performed using SAS (version 9.4, SAS Institute Inc., Cary, NC, USA), data analyzes were done using SPSS (version 21, IBM Corp, Armonk, NY, USA) software.

Factors associated with survival of the HCV patients on HD were analyzed using a univariate regression analysis and presented as Odds Ratios. A stepwise multivariate regression model was used to assess for factors that were independently associated with survival. The hazard ratio for not receiving HCV treatment adjusted for the factors independently associated with survival was calculated using a Cox regression model. A Kaplan-Meier survival curve was performed to visualize survival overtime and analyzed by a log-rank test. All reported *P*-values are two-sided.

REFERENCES

Büsch *et al.* Scand J Gastroenterol. 2016:1 Sep 6:1-8.
Büsch *et al.* EASL 2016:2 Abstract PS138
Chen and Morgan, Int. J. Med. Sci. 2006; 3(2): 47–52.
Fabrizi *et al.*, New J. Sci. New J. Sci. 2014 dx.doi.org/10.1155/2014/180203;
Gane E *et al.*, AASLD 2016 LB-11, Hepatology. 64:1125A;
Gilead, Sovaldi EMA SPC - (http://www.ema.europa.eu/docs/en_GB/document_library/EPAR_-_Product_Information/human/002798/WC500160597.pdf) [Accessed 19-Aug-2016]
Goodkin *et al.*, Am. J. Nephrol. 2013;38:405-412
Martin and Fabrizi, J. Hepatol. 2008 Oct;49(4):613-24.
Messina *et al.*, Hepatology 2015 Jan;61(1):77-87
Pockros *et al.* Gastroenterology 2016 (7):1590-98
Roth *et al.* The Lancet 2015 Oct 17;386(10003):1537-45.
SNR 2014. Svenska Njurregistret 2014 - (www.medscinet.net/snr/rapporterdocs/SNR%20%C3%85rsrapport%202014_Webb.pdf) [Accessed 19-aug-2016]
Thein *et al.* Hepatology 2008 Aug;48(2):418-31

RESULTS

CKD IN SWEDISH HCV PATIENTS

During 2001 and 2013, 2.5% (n=1,077) of the Swedish HCV patients had a reported CKD diagnosis (**Table 2**). This resulted in a higher incidence of CKD diagnosis within the HCV cohort than expected when comparing with the matched comparator cohort (3.84 cases per 1,000 person-years vs. 0.97 cases per 1,000 person-years in the HCV cohort and comparator cohort, respectively), resulting in a SIR of 4.0 (95% CI 3.7-4.2) (**Table 3**).

As previously presented the HCV patients had a higher risk of dying (SMR 5.6) (Büsch et al. 2016:2), which increased even further (SMR 6.9) in HCV patients with a CKD diagnosis (**Table 2**).

Table 3. Standard Incidence Ratio (SIR)

	Obs.	Exp.	SIR	95% CI
Acute kidney failure (N17[1])	845	139.1	6.1	5.7-6.5
Chronic kidney disease (N18[1])	1,077	270.7	4.0	3.7-4.2
Unspecified kidney failure (N19[1])	360	69.6	5.2	4.6-5.7
Kidney disease (N17[1]-N19[1])	1,815	387.0	4.7	4.5-4.9
Kidney cancer (180.0[2]/180.9[2])	41	24.0	1.8	1.2-2.2
Kidney transpl. (Z94.0[1])	340	49.3	6.9	6.2-7.6
Cryoglobulinemia (D89.1)[1]	47	0.4	129.1	92.2-166.0
Diabetes (E10-E14[1]/A10[3])	5,011	2,587.2	1.9	1.9-2.0

Obs. = Observed, Exp. = Expected, [1]ICD-10 codes, [2]ICD-7 codes, [3]ATC-codes

Table 2. Demographics and Disease Characteristics depending on CKD diagnosis or HD

	No CKD diagnosis (n=41,445)	CKD diagnosis (n=1,077)	Hemodialysis (n=268)
Person-years	273,426.2	6,696.6	1,816.1
Mean observation time (95% CI)	6.60 (6.56-6.64)	6.22 (5.98-6.46)	6.78 (6.35-7.20)
Dead	7,340 (18%)	559 (52%)	135 (50%)
SMR	5.6	6.9	10.4
Acute kidney failure (N17[1])	630 (1.5%)	215 (20%)	81 (30%)
Unspecified kidney failure (N19[1])	132 (0.1%)	228 (21%)	81 (30%)
Kidney disease (N17[1]-N19[1])	738 (1.8%	1,077 (100%)	265 (99%)
Kidney transplantation (Z94.0[1])	141 (0.3%)	199 (19%)	86 (32%)
Kidney cancer (180.0[2]/180.9[2])	35 (0.1%)	6 (0.6%)	3 (1%)
Diabetes (E10-E14[1]/A10[3])	116 (43%)	460 (43%)	116 (43%)
Gender (Male)	27,328 (66%)	744 (69%)	186 (69%)
<u>Marital status</u>			
Married	8,481 (20%)	403 (37%)	94 (35%)
Unmarried	23,342 (56%)	287 (36%)	55 (21%)
Divorced	8,872 (21%)	237 (22%)	108 (40%)
Widow	750 (1.8%)	50 (4.7%)	11 (4.1%)
<u>Country of origin</u>			
Sweden	33,177 (80%)	793 (74%)	178 (66%)
Europe	4,714 (11%)	159 (15%)	47 (18%)
Other	3,553 (8.6%)	125 (12%)	41 (16%)
Information missing	1 (<0.1%)	0	0
<u>Year of birth (grouped)</u>			
-1949	6,775 (16%)	513 (48%)	93 (35%)
1950-1969	22,936 (55%)	505 (47%)	152 (57%)
1970-1989	11,038 (27%)	55 (5.1%)	22 (8%)
1990-	696 (1.7%)	4 (0.4%)	1 (0.4%)

[1]ICD-10 codes, [2]ICD-7 codes, [3]ATC- codes

Figure 1. Cumulative Survival for HCV patients on HD grouped depending on IFN-based HCV treatment

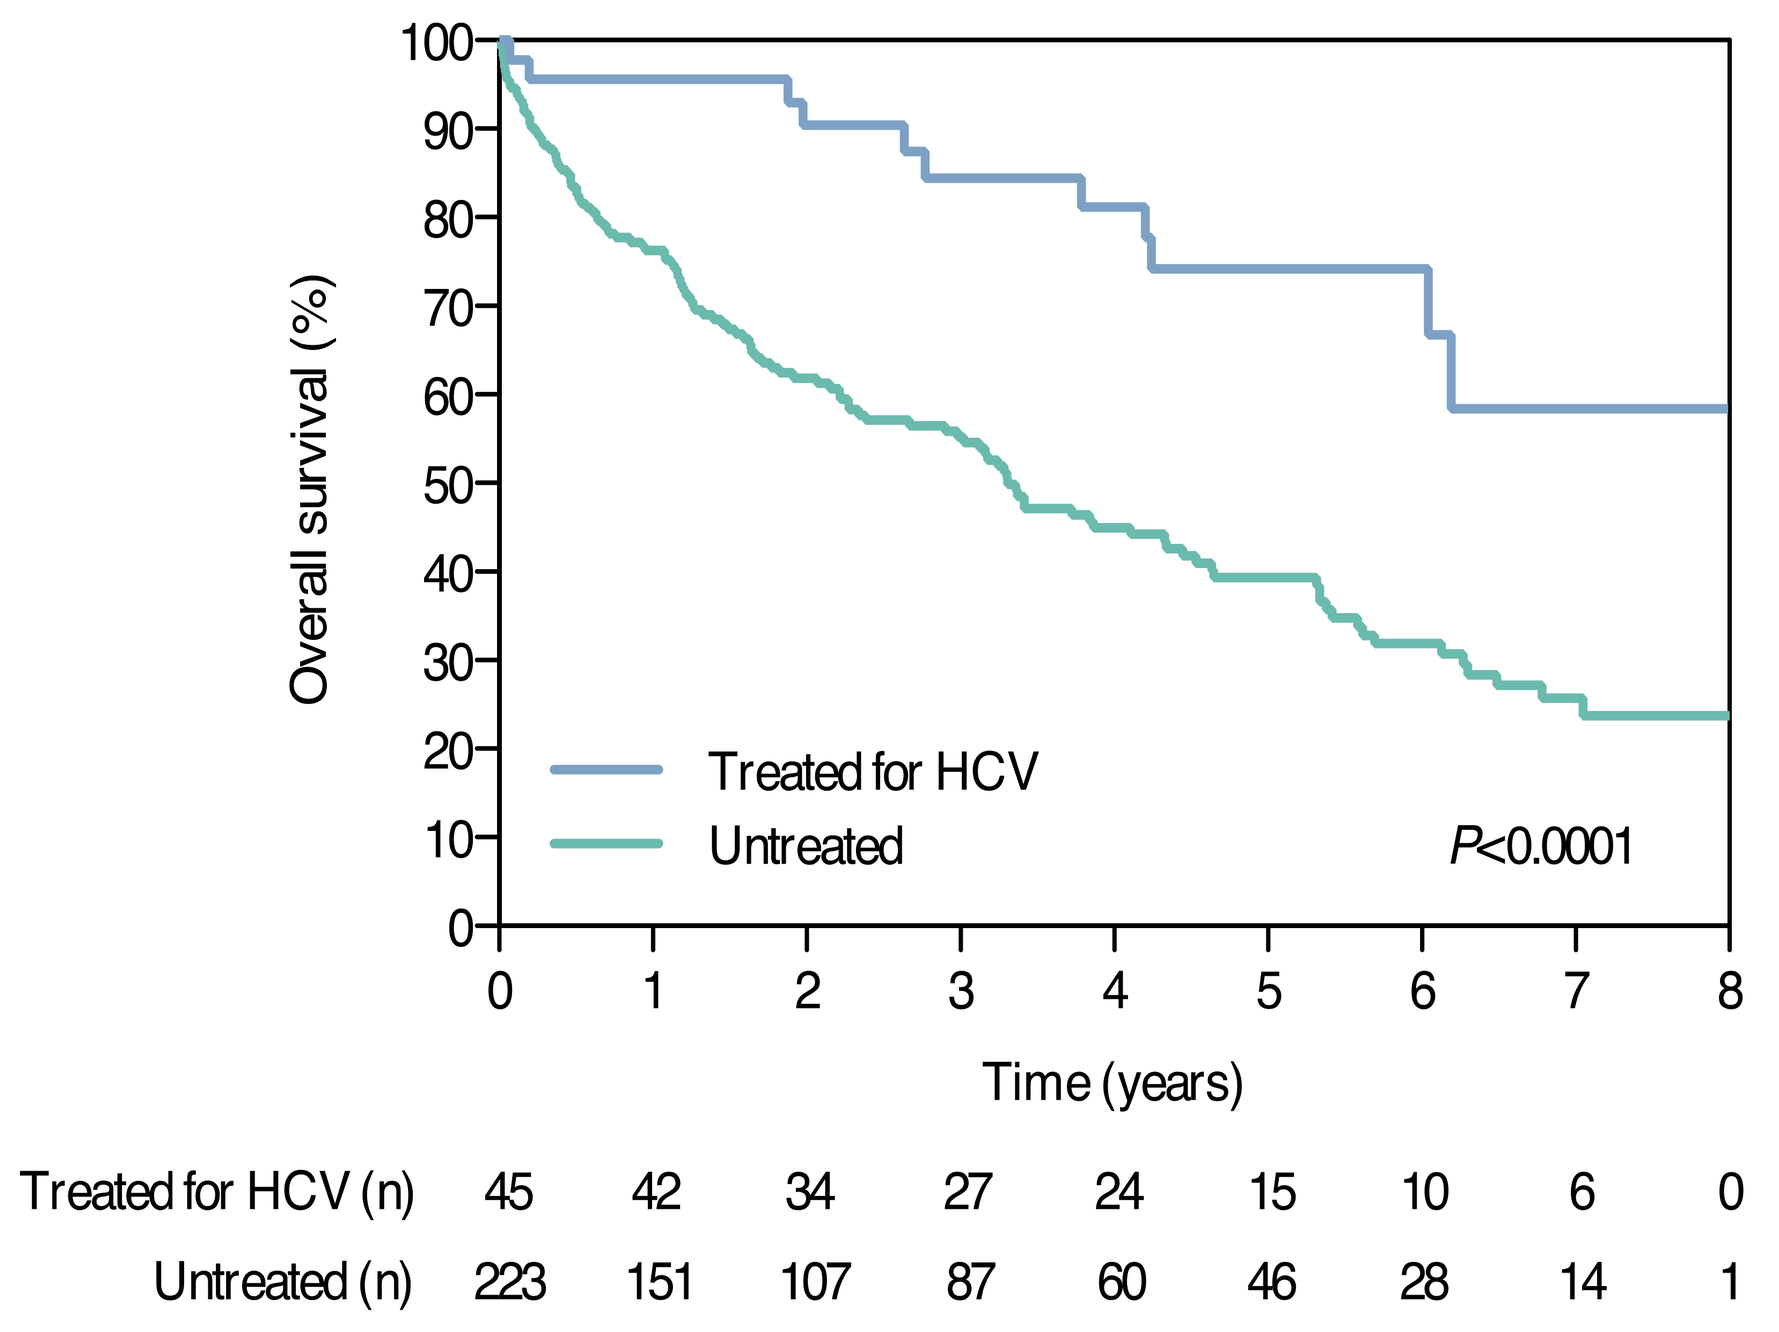
Overall survival (%)
100
90
80
70
60
50
40
30
20
10
0
Treated for HCV
Untreated
P<0.0001
0 1 2 3 4 5 6 7 8
Time (years)
Treated for HCV (n) 45 42 34 27 24 15 10 6 0
Untreated (n) 223 151 107 87 60 46 28 14 1

HEMODIALYSIS

In total 268 HCV patients had ≥1 HD procedure code during the study period. Of those, 50% had died with resulting SMR of 10.4 for the cohort (**Table 2**). Almost all HD patients were accompanied with kidney failure diagnosis. The SUR for receiving HD was 7.0 (95% CI 6.2-7.9).

Younger age at initiating HD, receiving either kidney transplantation or HCV treatment, or an acute kidney failure diagnosis was significantly associated with survival in hemodialysis patients when analyzing using a univariate regression model. All four factors also independently predicted survival when using a multivariate regression analysis (**Table 4**).

- The overall survival in HCV patients on HD was significantly higher if receiving HCV treatment (**Figure 1**). N.B. no information on treatment outcome was available in the registry.
- The mortality hazard ratio for not receiving HCV treatment after adjusting for kidney transplantation, age, and acute kidney failure diagnosis was 2.874 (1.541-5.358, P=0.001).
- The annual prevalence of HCV among hemodialysis patients in Sweden was between 3% and 4% for the years 2010 through 2013. The overall chronic HCV prevalence in Sweden is estimated to be around 0.36%.
- Of the HD patients 17% (45/268) had received HCV treatment, 60% (27/45) of these received HCV treatment after initiating HD – i.e. 10% (27/268) started treatment after HD initiation.

Table 4. Univariate and Multivariate Analysis of Factors associated with Survival in HCV patients on hemodialysis (Odds Ratios)

	Univariate	95% CI	*P*-value	Multivariate	95% CI	*P*-value
Acute kidney failure (N17[1])	2.611	1.412-4.829	**0.002**	2.518	1.394-4.547	**0.002**
Chronic kidney disease (N18[1])	0.877	0.237-3.244	0.8			
Unspecified kidney failure (N19[1])	0.655	0.358-1.200	0.2			
Kidney transpl. (Z94.0[1])	2.720	1.472-5.026	**0.001**	2.976	1.649-5.379	**0.0003**
Kidney cancer (180.0[2]/180.9[2])	2.196	0.189-25.528	0.5			
Cryoglobulinemia (D89.1)[1]	0.390	0.020-7.593	0.5			
Diabetes (E10-E14[1]/A10[3])	0.941	0.543-1.633	0.8			
Gender (Male)	1.329	0.729-8.850	0.3			
HCV treatment (n=45)	3.901	1.720-8.850	**0.001**	3.547	1.634-7.793	**0.001**
Age at HD initiation	0.965	0.942-0.989	**0.004**	0.966	0.943-0.989	**0.004**

CONCLUSIONS

- There was an increased risk for CKD diagnosis among HCV patients
- Internationally, Sweden has a high HCV treatment rate of HD patients
- Younger age at initiating HD, receiving either a kidney transplantation or HCV treatment, or having an acute kidney failure diagnosis in patients requiring HD were independently associated with having a better survival
- The adjusted increased risk of dying in HD patients that never received HCV treatment was 287%. This result may suggest that anti-viral treatment during HD may confer better survival for patients waitlisted for transplantation.

DISCLOSURES

JS, JK, and KB are employees of AbbVie and may hold stocks or stock options. RS honorairies for lectures from Abbvie, MSD/Merck, Medivir, and BMS. AB consulting fees from ChemoCentryx and Merck/MSD, and honoraries for lectures from Abbvie, Chemocentryx, Merck/MSD, Sanofi-Genzyme. KL honorairies for lectures from Abbvie, Merck/MSD, Medivir, Gilead, BMS. CM has nothing to disclose.

The design, study conduct, and financial support for the study were provided by AbbVie. AbbVie participated in the study design, data analysis, interpretation of results, review and approval of the publication. The authors determined the final content. No payments were made to the authors for writing this publication.

54 ERA
305--SP
CDK - Clinical epidemiology I
Annette Bruchfeld
DOI: 10.3252/pso.eu.54ERA.2017
ePosters supported by F. Hoffmann- La Roche Ltd.