SP-124

In Vitro Evaluation Induction of Cytochrome P450 Enzymes by Sparsentan and Effects on Its Metabolism by Potential Co-Administered Drugs

Kevin Leach, PhD and Xin-Ru Pan-Zhou, PhD

Retrophin, Inc., Cambridge, MA

Introduction

Sparsentan (RE-021) is a first-in-class, dual endothelin A and angiotensin 2 type 1 receptor antagonist that has been evaluated in a clinical trial (DUET) for the treatment of primary focal segmental glomerulosclerosis (FSGS), a serious kidney disorder that when left untreated often leads to end-stage renal disease (**Figure 1**). Sparsentan is primarily metabolized by CYP3A4 and is not a potent inhibitor of the major human CYPs in vitro (Pan-Zhou et al, ASN 2016). Sparsentan was slowly turned over in human microsomes and in plasma samples from FSGS patients, sparsentan was the major drug-related component in circulation, representing >85% of the total drug-related material. Each individual metabolite accounted for <6% of the total material.

Objective

CYP Induction Potential: to investigate the effects of sparsentan on the mRNA expression and enzyme activities (CYP1A2, CYP2B6 and CYP3A4) in primary cultures of human hepatocytes.

CYP DDI Potential: to assess the potential for drugs that may be co-administered due to their use in FSGS with sparsentan including cyclosporine A, fluvastatin, fenofibrate, and voriconazole as a CYP3A inhibitor control to inhibit the metabolism of sparsentan in human liver microsomes. Lastly, to determine the rates of sparsentan metabolism in human hepatocytes treated for 72 hours with CYP inducers known to be used by patients with FSGS, including cyclosporine A, prednisone, torsemide, atorvastatin, cerivastatin, fluvastatin, pravastatin, and simvastatin.

Figure 1. Sparsentan Structure

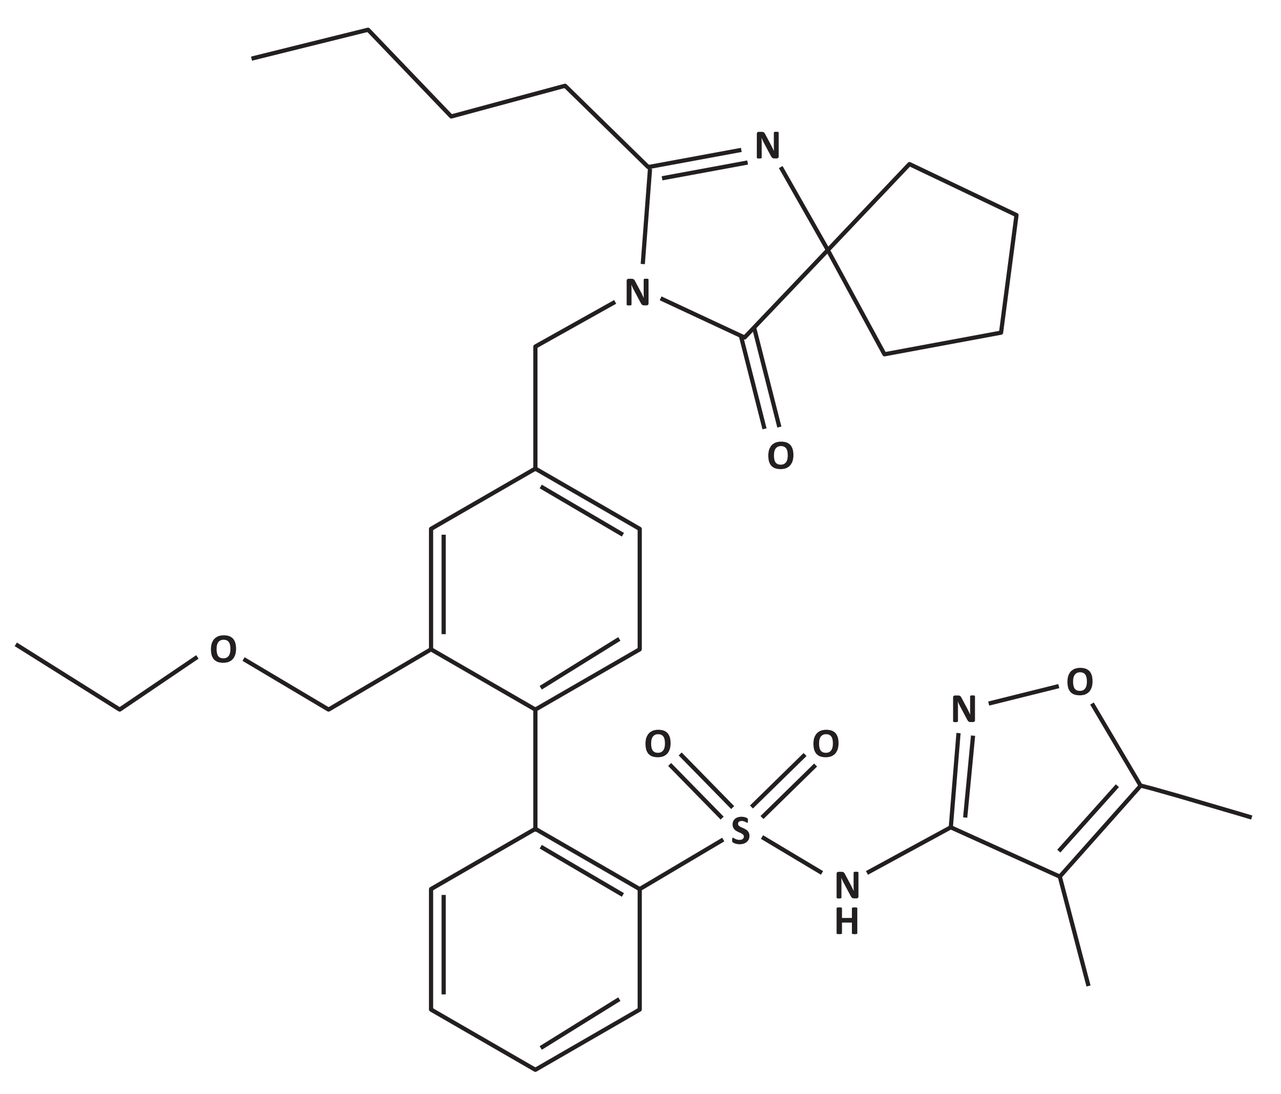

4'-[(2-Butyl-4-oxo-1,3-diazaspiro[4.4]non-1-en-3-yl)methyl]-N-(4,5-dimethyl-3-isoxazolyl)-2'-(ethoxymethyl)-[1,1'-biphenyl]-2-sulfonamide

Methods

CYP Induction Potential

- Cytotoxicity evaluation for sparsentan (3, 10, 30, 100, 250 or 550 µM) or clotrimazole (10, 60 or 100 µM, positive control) was assessed using the lactate dehydrogenase assay (a measure of cell membrane integrity) by measurement of the reduction of resazurin (a measure of mitochondrial respiration) in human hepatocytes treated for 3 consecutive days.
- CYP1A2, CYP2B6 and CYP3A4 induction was assayed with hepatocytes incubated with sparsentan (1, 5, 10, 30, 100 or 250 µM) for 3 days, followed either by measurement of mRNA for the expression levels of the CYP genes or by the measure of CYP activities using marker substrates (phenacetin, bupropion, or testosterone) for phenacetin O-dealkylase (CYP1A2), bupropion hydroxylase (CYP2B6), and testosterone 6β-hydroxylase (CYP3A4/5) activities.

CYP DDI Potential of Sparsentan

- DDI inhibition potential was conducted using pooled human liver microsomes (0.5 mg/mL) incubated with 5 µM sparsentan in the presence or absence of cyclosporine A, fluvastatin, fenofibrate, or voriconazole (control CYP3A inhibitor) for 30 minutes. The concentrations of these likely co-administered drugs were 1-, 3-, and 10-fold plasma C_{max}. The inhibition of sparsentan was assessed by measurement of the disappearance of sparsentan as well in the same experiment, as the formation of sparsentan metabolites (one O-deethylated and five different hydroxylated metabolites).
- DDI induction potential was conducted using human hepatocytes treated with rifampin, an inducer of CYP3A4 and a non-inducer (flumazenil) as well as potential co-administered drugs prepared at a final acetonitrile or methanol concentration of ≤1.0% (v/v) in medium for 72 h (**Table 1**). The medium was refreshed every 24 h. At the end of the incubation period, the medium was removed and replaced with medium containing CYP3A4 substrate (testosterone, 250 µM) or sparsentan (5 µM). Incubations of sparsentan were terminated with acetonitrile after 0, 1, 2 or 4 h whereas testosterone incubation was only for 2 h. Incubations were terminated after 2 hours. CYP3A4 activity was determined by quantitating the CYP3A4 selective metabolite (6β-hydroxytestosterone). The sparsentan metabolism rate was determined by quantitating remaining sparsentan by LC-MS/MS.

Table 1. Compounds Tested for DDI Induction Potential With Sparsentan

Treatment	Concentration (µM)
Vehicle[a]	Not applicable
Rifampicin	20
Flumazenil	20
Cyclosporine A	0.8 and 8
Prednisone	0.03 and 0.3
Torsemide	4.2 and 42
Atorvastatin	0.12 and 1.2
Cerivastatin	0.03 and 0.3
Fluvastatin	1.1 and 11
Pravastatin	0.07 and 0.7
Simvastatin	0.02 and 0.2

Cell culture medium contained 1% acetonitrile or 1% methanol. The vehicle control for all treatments was 1% acetonitrile except the vehicle control for atorvastatin and torsemide was 1% methanol.

Results

Table 2. CYP1A2, CYP2B6, and CYP3A4 Catalytic Activity Fold Changes in Sparsentan/Positive/Negative Treated Cultured Human Hepatocytes

	CYP1A2 (Phenacetin O-dealkylation) Fold Change			CYP2B6 (Bupropion hydroxylation) Fold Change			CYP3A4 (Testosterone 6β-hydroxylation) Fold Change		
Sparsentan Concentration (µM)	Donor 1	Donor 2	Donor 3	Donor 1	Donor 2	Donor 3	Donor 1	Donor 2	Donor 3
0.1% DMSO	1	1	1	1	1	1	1	1	1
1	0.82	1.20	1.18	0.91	1.59	1.63	1.64	2.63	2.31
5	0.74	1.15	1.44	1.52	2.29	3.39	2.7	2.91	5.87
10	1.04	1.07	1.49	1.63	2.60	7.00	1.19	2.63	4.57
30	1.32	1.2	1.69	2.97	3.77	7.03	0.58	1.47	2.54
100	1.75	0.95	2.30	2.58	3.47	7.41	0.44	1.20	1.89
250	0.94	1.08	1.36	1.22	2.07	2.50	0.18	0.27	0.64
Positive Control	Omeprazole (50 µM)			Phenobarbital (750µM)			Rifampin (20µM)		
	40.3	41.5	73.7	9.99	12.2	23.9	7.99	25.6	52.2
Negative Control	Flumazenil (25µM)			Flumazenil (25µM)			Flumazenil (25µM)		
	0.84	1.17	1.04	1.01	1.05	0.88	0.94	1.02	1.03

Table 3. CYP1A2, CYP2B6, and CYP3A4 mRNA Fold Changes in Sparsentan/Positive/Negative-Treated Cultured Human Hepatocytes

	CYP1A2 mRNA Fold Change			CYP2B6 mRNA Fold Change			CYP3A4 mRNA Fold Change		
Sparsentan Concentration (µM)	Donor 1	Donor 2	Donor 3	Donor 1	Donor 2	Donor 3	Donor 1	Donor 2	Donor 3
0.1% DMSO	1	1	1	1	1	1	1	1	1
1	0.90	1.05	1.05	1.22	1.63	1.06	2.92	3.96	3.28
5	0.84	1.15	1.03	1.55	2.67	1.63	6.80	10.1	16.6
10	0.76	0.84	0.95	1.93	2.75	2.26	7.61	10.5	19.3
30	0.70	0.57	0.77	2.12	2.37	2.80	8.23	9.00	17.7
100	1.02	0.74	0.96	2.58	2.78	3.15	8.28	10.9	20.2
250	1.16	0.70	0.63	2.23	2.54	2.12	5.19	4.78	8.62
Positive Control	Omeprazole (50 µM)			Phenobarbital (750µM)			Rifampin (20µM)		
	55.8	64.1	40.4	7.97	9.82	6.33	12.4	27.2	48.8
Negative Control	Flumazenil (25µM)			Flumazenil (25µM)			Flumazenil (25µM)		
	1.03	0.99	1.06	0.85	1.04	0.99	1.09	1.07	1.01

- Sparsentan had little or no effect (i.e. <2.0-fold change and/or <20% of the positive control) on CYP1A2 activity and mRNA levels.
- Sparsentan caused concentration-dependent increases in CYP2B6 activity which reached 2.97- to 7.41-fold change and mRNA levels which reached 2.58- to 3.15-fold change and >20% of the positive control in all 3 hepatocyte preparations for both mRNA and activity (**Table 2** and **Table 3**).
- Sparsentan caused increases between 2.70- and 5.87-fold change in CYP3A4/5 activity at lower concentrations (1 and/or 5 µM) and were >20% of the positive control in one of the 3 hepatocyte preparations (**Table 2**).
- Sparsentan caused increases in CYP3A4 mRNA levels which reached 8.28- to 20.2-fold change and were >20% of the positive control in all 3 hepatocyte preparations, although these increases were not concentration-dependent in two of 3 hepatocyte preparations (**Table 3**).

Figure 2. Evaluation of Potential Co-Administrated Drugs on Sparsentan Metabolism

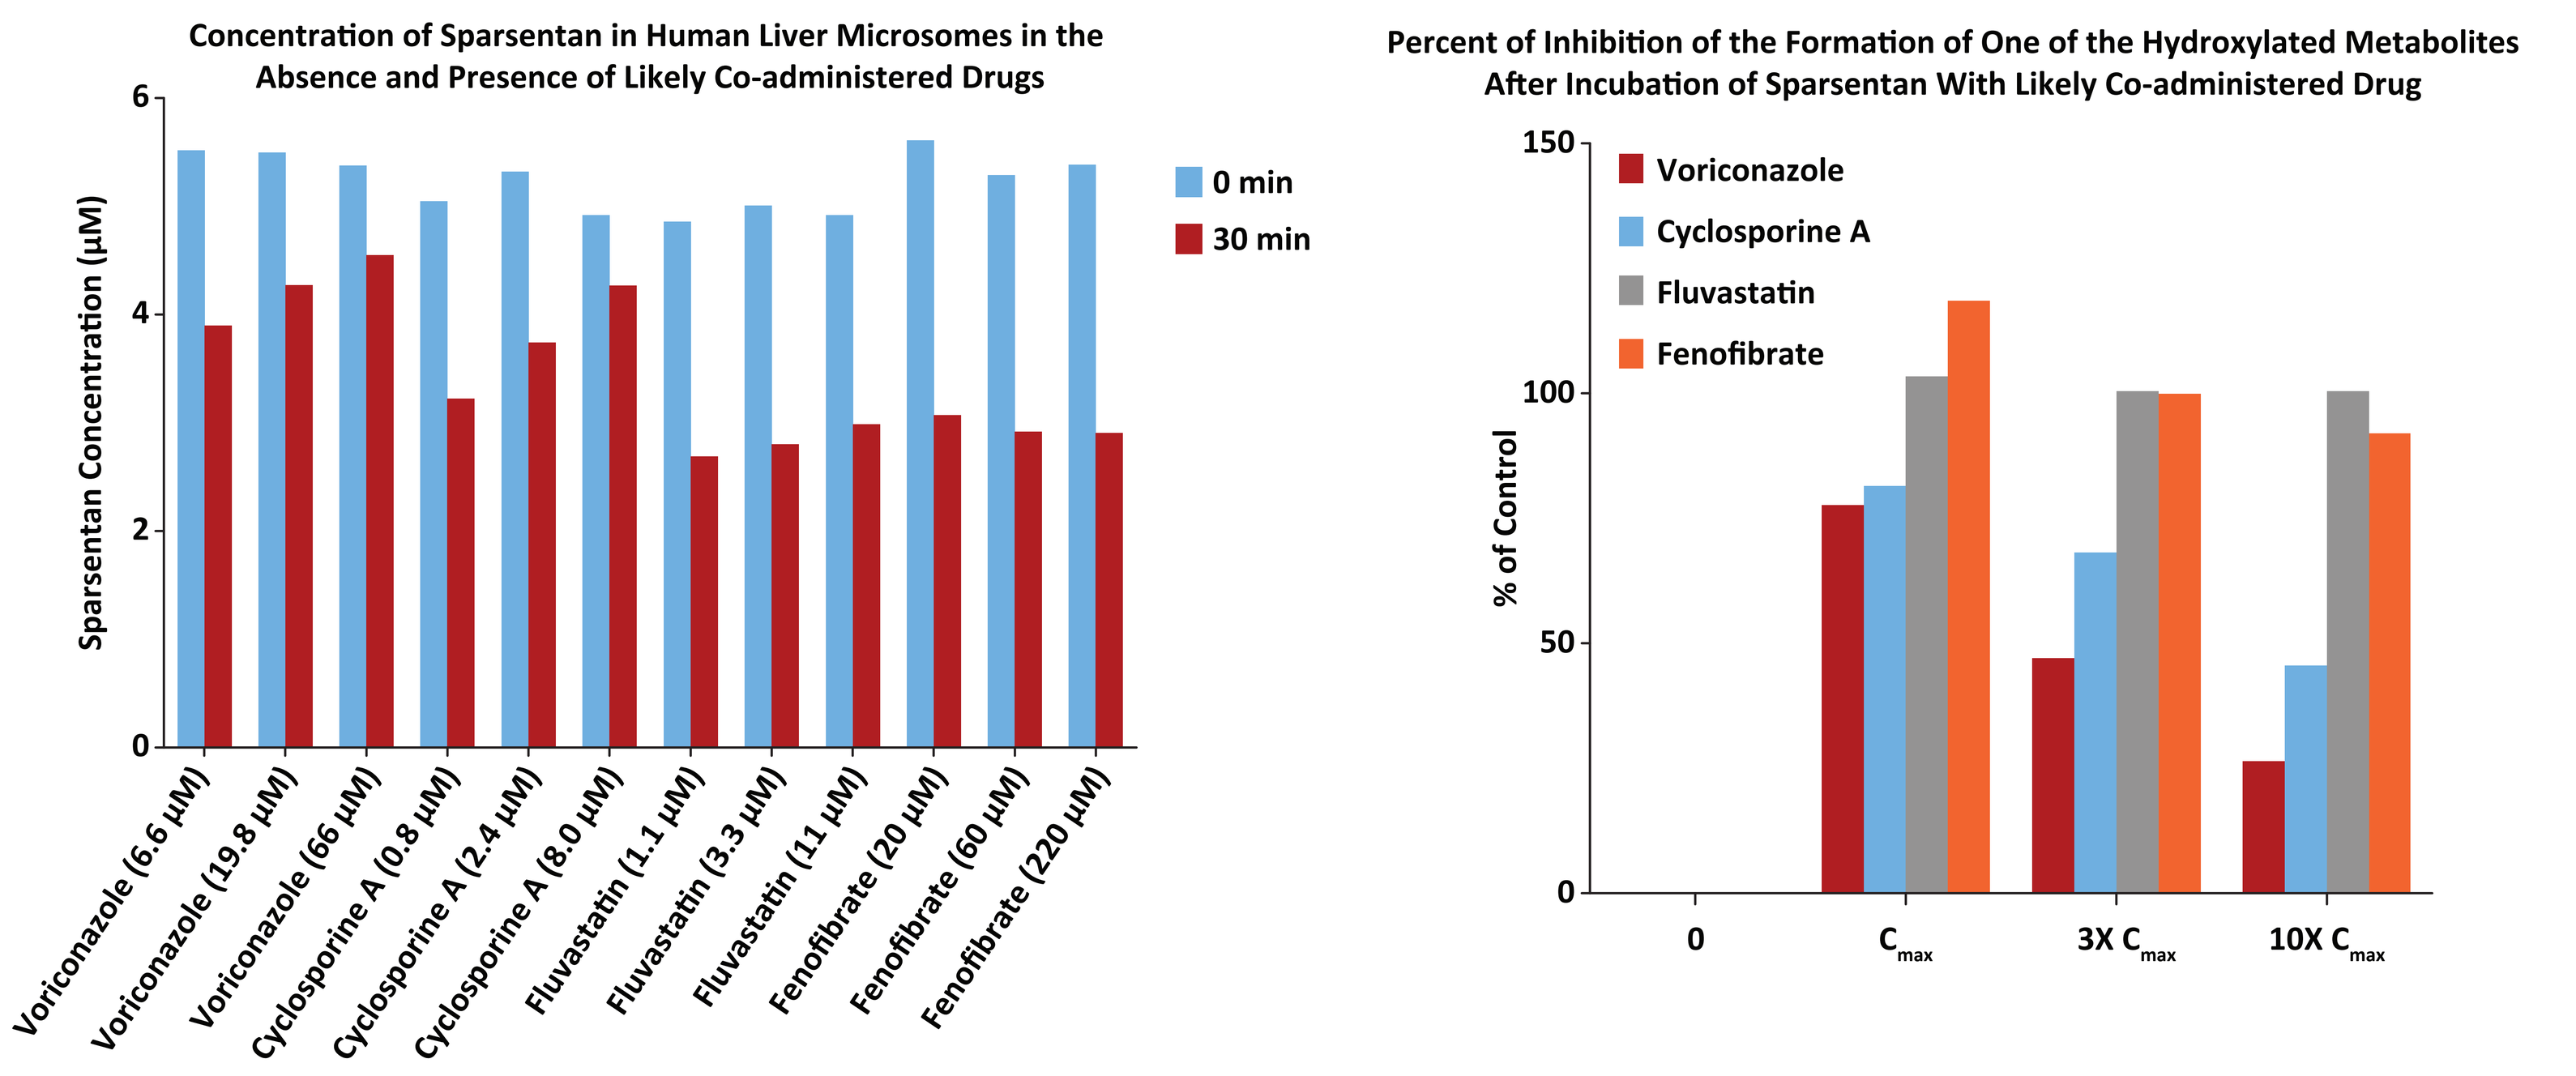
Concentration of Sparsentan in Human Liver Microsomes in the Absence and Presence of Likely Co-administered Drugs
Sparsentan Concentration (µM)
0 min
30 min
Voriconazole (6.6 µM)
Voriconazole (19.8 µM)
Voriconazole (66 µM)
Cyclosporine A (0.8 µM)
Cyclosporine A (2.4 µM)
Cyclosporine A (8.0 µM)
Fluvastatin (1.1 µM)
Fluvastatin (3.3 µM)
Fluvastatin (11 µM)
Fenofibrate (20 µM)
Fenofibrate (60 µM)
Fenofibrate (220 µM)
Percent of Inhibition of the Formation of One of the Hydroxylated Metabolites After Incubation of Sparsentan With Likely Co-administered Drug
% of Control
Voriconazole
Cyclosporine A
Fluvastatin
Fenofibrate
0
C_{max}
3X C_{max}
10X C_{max}

- Cyclosporine A and voriconazole, but not fluvastatin and fenofibrate, showed concentration-dependent inhibition of sparsentan metabolism (**Figure 2**).

Figure 3. Evaluation of CYP3A4 Induction Effect on Sparsentan Metabolism

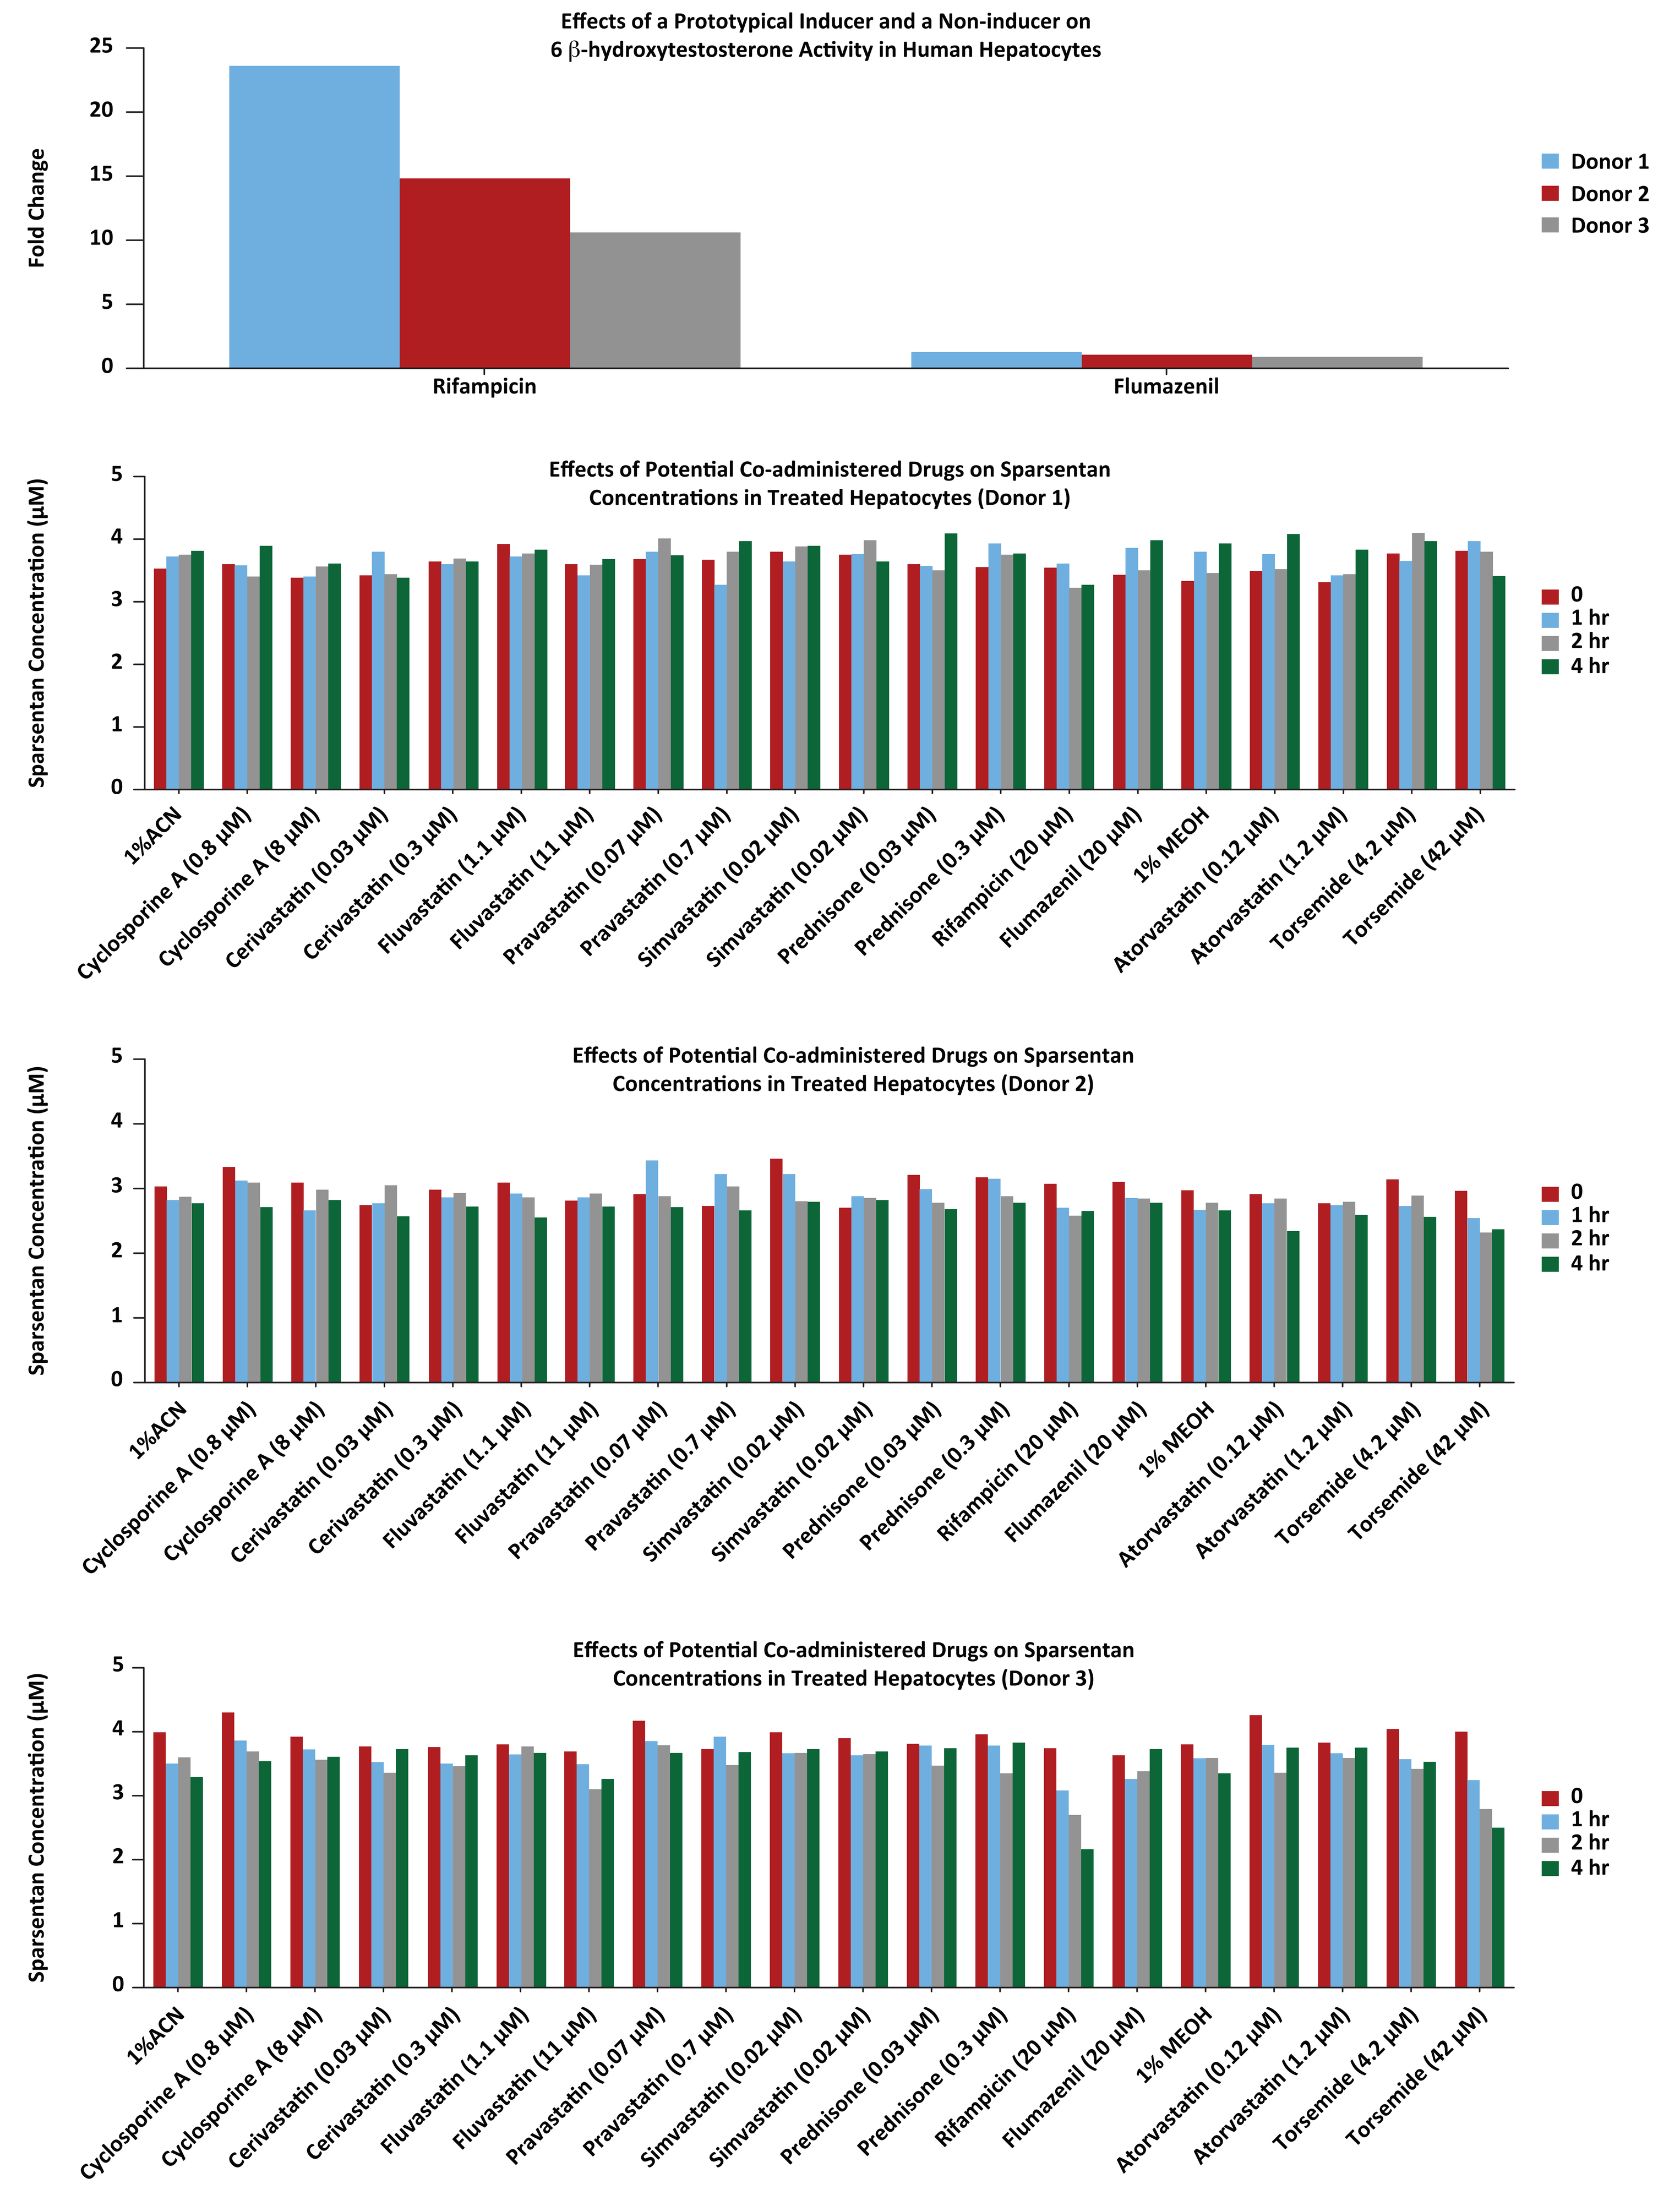
Effects of a Prototypical Inducer and a Non-inducer on 6 β-hydroxytestosterone Activity in Human Hepatocytes
Fold Change
Rifampicin
Flumazenil
Donor 1
Donor 2
Donor 3
Effects of Potential Co-administered Drugs on Sparsentan Concentrations in Treated Hepatocytes (Donor 1)
Effects of Potential Co-administered Drugs on Sparsentan Concentrations in Treated Hepatocytes (Donor 2)
Effects of Potential Co-administered Drugs on Sparsentan Concentrations in Treated Hepatocytes (Donor 3)
Sparsentan Concentration (µM)
0
1 hr
2 hr
4 hr
1%ACN
Cyclosporine A (0.8 µM)
Cyclosporine A (8 µM)
Cerivastatin (0.03 µM)
Cerivastatin (0.3 µM)
Fluvastatin (1.1 µM)
Fluvastatin (11 µM)
Pravastatin (0.07 µM)
Pravastatin (0.7 µM)
Simvastatin (0.02 µM)
Simvastatin (0.02 µM)
Prednisone (0.03 µM)
Prednisone (0.3 µM)
Rifampicin (20 µM)
Flumazenil (20 µM)
1% MEOH
Atorvastatin (0.12 µM)
Atorvastatin (1.2 µM)
Torsemide (4.2 µM)
Torsemide (42 µM)

- The metabolism of testosterone, a selective CYP3A4 substrate, was increased after incubation for 2 hours with hepatocytes treated with rifampicin, flumazenil, or 1% acetonitrile. These results indicate that the hepatocytes used in the study were competent for CYP3A4 induction (**Figure 3**, top bar chart).
- Among 1 out 3 donors, the metabolism of sparsentan was enhanced (remaining sparsentan <85% of the vehicle control) in hepatocytes treated with rifampicin at 20 µM and torsemide at 42 µM (10xC_{max}), but not at 4.2 µM.
- No effects were observed on sparsentan metabolism in hepatocytes treated by cyclosporine A, prednisone, simvastatin, atorvastatin, cerivastatin, fluvastatin, pravastatin, and flumazenil in all 3 donors.

Conclusions

- The *in vitro* studies indicated sparsentan is not CYP1A2 inducer but may induce CYP2B6 and CYP3A4, further use of mechanistic models to determine the degree of drug-drug interactions may be warranted for further *in vivo* evaluation.
- Cyclosporine A and voriconazole, but not fluvastatin and fenofibrate, showed concentration-dependent inhibition of sparsentan metabolism, indicating the need for *in vivo* evaluation of sparsentan exposure with potential CYP3A4 potent inhibitors.
- The metabolism of sparsentan can be enhanced in human hepatocytes treated with rifampicin and torsemide.
- Phase 1 clinical drug-drug interaction studies are underway using some of the same probes used in the *in vitro* studies to define the potential for DDIs with co-administered medications (**Figures 4** and **5**).

Figure 4. CYP3A4 and CYP2B6 Induction DDI Study Design

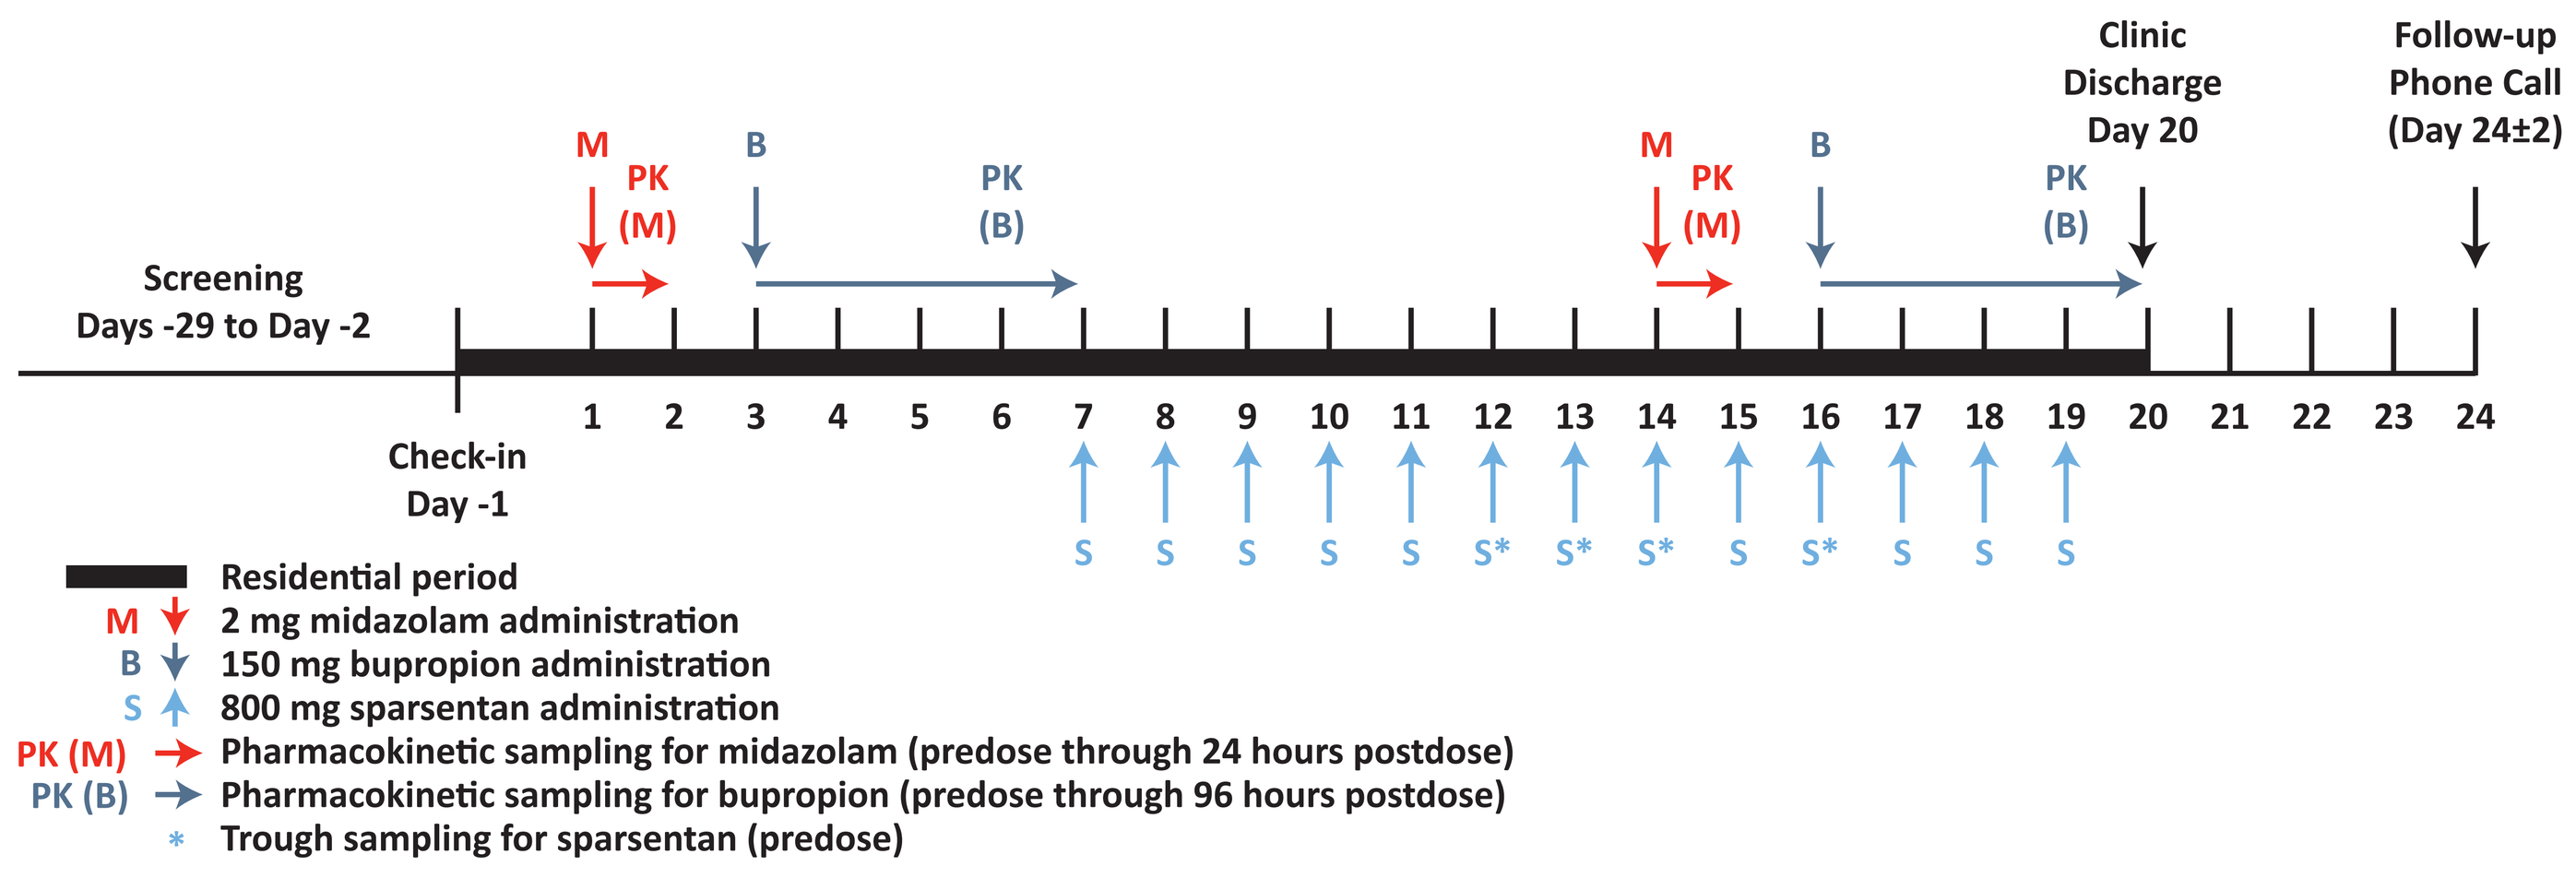
Clinic Discharge Day 20
Follow-up Phone Call (Day 24±2)
M
PK (M)
B
PK (B)
Screening Days -29 to Day -2
Check-in Day -1
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24
S S S S S S* S* S* S S* S S S
Residential period
M 2 mg midazolam administration
B 150 mg bupropion administration
S 800 mg sparsentan administration
PK (M) Pharmacokinetic sampling for midazolam (predose through 24 hours postdose)
PK (B) Pharmacokinetic sampling for bupropion (predose through 96 hours postdose)
* Trough sampling for sparsentan (predose)

Figure 5. CYP3A4 Inhibition DDI Study Design

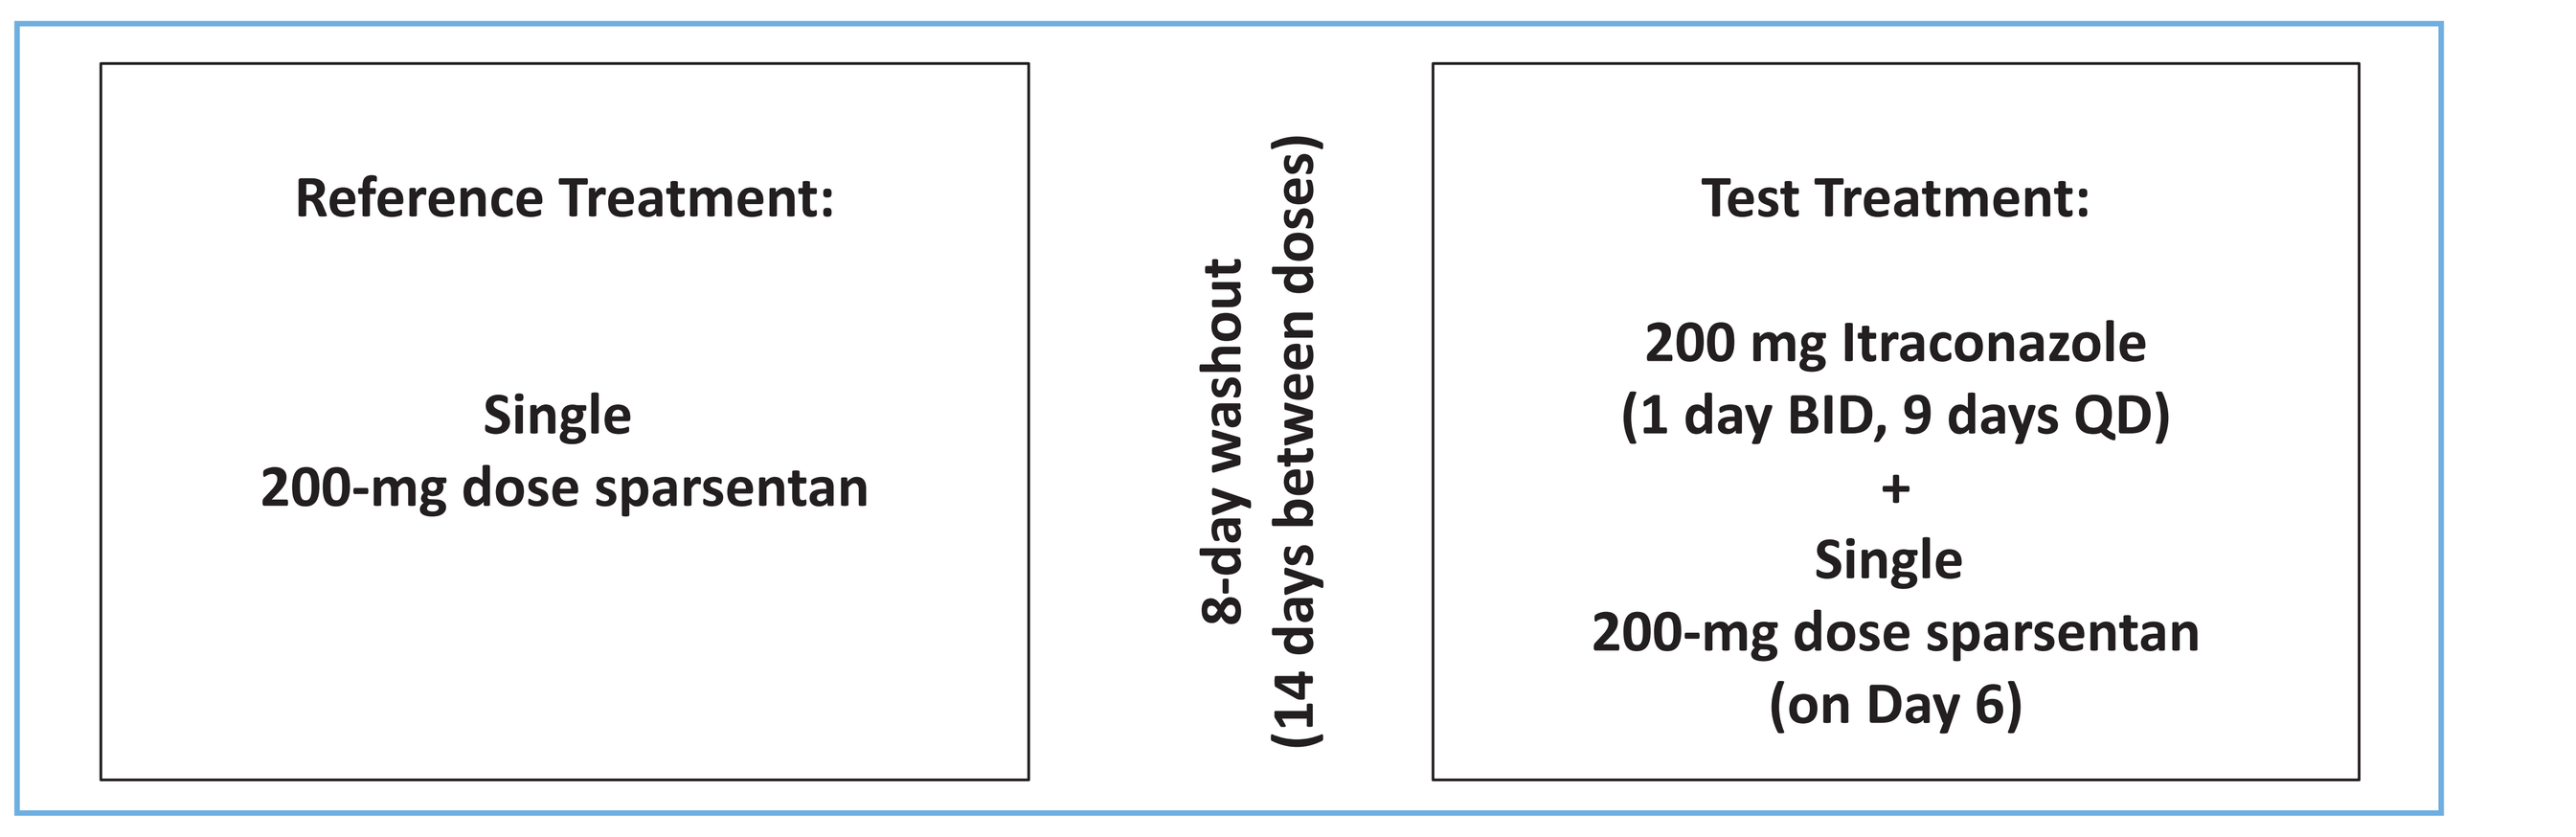
Reference Treatment:
Single
200-mg dose sparsentan
8-day washout
(14 days between doses)
Test Treatment:
200 mg Itraconazole
(1 day BID, 9 days QD)
+
Single
200-mg dose sparsentan
(on Day 6)

Reference

Pharmacokinetics of Sparsentan in Healthy Subjects: In Vitro Metabolism and Effects of Food, Gender, Age, and Multiple-Dose Escalation. Xin-Ru Pan-Zhou, Kevin Leach and Maria Beconi. Retrophin, Inc., Cambridge, MA, Presented at the American Society of Nephrology Kidney Week, 15–20 November 2016, Chicago, Illinois, USA

Acknowledgment

The authors thank Dr. Luo Gang at Covance and Mr. Paul Bolliger at Xenotech for their *in vitro* metabolism work.

Disclosure Information

Xin-Ru Pan-Zhou and Kevin Leach are full-time employees of Retrophin, Inc., and own stock/stock options. The company is developing sparsentan for potential treatment of primary FSGS.

Retrophin, Inc. acquired sparsentan from Ligand Pharmaceuticals, Inc. (formerly Pharmacopeia). Retrophin funded the *in vitro* metabolism studies. Retrophin participated in the study design, data collection and interpretation, writing, reviewing, and approving this abstract for submission.

All authors had access to the data; participated in the development, review, and approval of the poster during the development process. Poster graphical and editorial support was provided by Richard Perry, PharmD and was funded by Retrophin.

Presented at ERA-EDTA, 3-6 June 2017, Madrid, Spain

54 ERA
124--SP
Glomerulonephritis I
Xin-ru Pan-Zhou
DOI: 10.3252/pso.eu.54ERA.2017
ePosters supported by F. Hoffmann- La Roche Ltd.