Serum C3 and Renal Outcome in Patients with Primary Focal Segmental Glomerulosclerosis

Jian Liu, JingyuanXie, Xiaoyan Zhang, Jun Tong, Xu Hao, Hong Ren, Weiming Wang, Nan Chen

Department of Nephrology, RuiJin Hospital, Shanghai Jiao Tong University, School of Medicine, Shanghai, China

Institute of Nephrology, Shanghai Jiao Tong University School of Medicine, Shanghai, China

Aim： The role of complement in the pathogenesis or progression of FSGS is uncertain. The aim of this observational cohort study was to identify the clinical implications of serum C3 levels and to investigate their utility as predictor of renal outcomes in patients with FSGS.

Methods： 591 biopsy-proven primary FSGS patients were recruited. Clinical, histological and progression data were recorded. Decreased serum C3 level was defined as C3 <85 mg/dl. The study endpoint was end-stage renal disease (ESRD).

Results： Of the patients, there were 117 patients (25.1%) with low serum C3. When renal biopsy was taken, compared to patients with C3 ≥85 mg/dL, those with C3 <90 mg/dL had higher level of serum creatinine, lower levels of eGFR, hemoglobin, proteinuria, triglyceride, cholesterol, IgA, more severe tubulointerstitial injury(TI). Multivariate linear regression analysis showed low C3 level was an independent risk factor for lower eGFR (HR=42.56, 95%CI 11.21- 73.91, p<0.01) after adjusted by sex, age and clinical parameters. Of 221 patients with a mean follow-up of 53.3 months, ESRD occurred in 32 patients (37.2%) with low serum C3 compared with 22 patients (16.3%) with normal C3 levels (P <0.001). Serum C3 level had a significant predictive value for renal outcome (AUC = 0.650, P = 0.001), The risk of reaching ESRD was significantly higher in patients with low serum C3 level (HR 4.044; 95% CI= 2.238 to 7.309; P < 0.001). Further, we included 40 primary FSGS and found significant correlation between MAC and AP (p=0.003), MAC and serum C3(p=0.018), AP and serum C3(p=0.028). Compared with patients with none to mild TI, those with moderate to severe TI tend to have lower level of serum C3(88.38±18.899 vs 87.38±15.794 mg/dL), lower level of AP (44.57±32.71 vs 46.50±30.85%) and higher level of serum MAC (3646.72±1573.09 vs 3431.02±1334.34ng/dL).

Conclusion： Complement activation occurs in patients with FSGS. It is associated with clinical and histological severities. Low serum C3 was an independent risk factor for the poor renal outcome in patients with FSGS.

SP-109

54TH ERA-EDTA CONGRESS
Madrid, Spain
June 3rd-6th 2017

54 ERA 109--SP

Glomerulonephritis I

Jian LIU

DOI: 10.3252/pso.eu.54ERA.2017

ePosters supported by F. Hoffmann- La Roche Ltd.

PosterSessionOnline.com