PRECLINICAL AND CLINICAL VALIDATION OF A NOVEL PHARMACO-DYNAMIC ASSAY TO EVALUATE THE EFFECT OF CALCIFICATION INHIBITORS ON CALCIUM PHOSPHATE CRYSTALLIZATION IN PLASMA

M.D. Ferrer[1,2], M.M. Pérez[1], M. Lazo[3], E. Bañón[3], J.M. Campistol[3], R. Ojeda[3], P.H. Joubert[1,4], M. Arias[3], AZ. Canals[1], J.V. Torregrosa[3], F. Maduell[3], C. Salcedo[1], J. Perelló[1,5]

[1] Laboratoris Sanifit SL., Palma, Spain; [2] Dep. Biologia Fonamental i C. de la Salut, University of the Balearic Islands (UIB), Palma, Spain; [3] Hospital Clínic, Nephrology and Renal Transplantation, Barcelona, Spain; [4] King's College, London, UK; [5] Laboratori d'Investigació en Litiasi Renal, UIB, Palma, Spain

Contact: joan.perello@sanifit.com

INTRODUCTION

Cardiovascular calcification (CVC) is a progressive complication of end-stage renal disease (ESRD) and is a predictor of cardiovascular events and mortality. The use of biomarkers to predict cardiovascular risk and the impact of treatment could have a crucial impact on the management of CVC. SNF472 is a new calcification inhibitor being developed by Sanifit for the management of calciphylaxis and CVC in patients with ESRD undergoing hemodialysis (HD).

AIM

To develop and validate a new Pharmacodynamic (PD) assay to measure the crystallization rate in rat and human plasma.

MATERIALS AND METHODS

PD Assay: This is a spectrophotometric assay performed in 96-well plates. Plasma was centrifuged at 10,000 g for 30 min and mixed with hydrogen phosphate and calcium to attain final concentrations of 1.5 mM phosphate and 12.5 mM calcium. Crystallization of calcium phosphate was monitored for 30 min by assessing the increase in sample turbidity. The plate was incubated at room temperature with continuous agitation in an orbital shaker. Plasma crystallization potential was assessed based on the slopes of the linear range from plots of increase in absorbance versus logarithm of time. The efficacy of calcification inhibitors was determined comparing plasma samples with and without the addition of inhibitors. The assay was fully validated analytically for SNF472.

Preclinical validation: Validation was performed using a rat model of calcification induced by subcutaneous administration of 75,000 IU/kg vitamin D for three consecutive days.

Clinical validation: Samples for the PD assay were collected from a double-blind, randomized, repeated dose study in HD patients in which SNF472 was administered by a 4-hour infusion during dialysis sessions. Eight subjects participated in a Multiple Ascending Dose (MAD) study, consisting of five one week Treatment Periods, with a 3-week washout period between doses. The doses were: 1, 3, 5, 12.5 and 20 mg/kg.

RESULTS

PD Assay: The in vitro addition of several different calcification inhibitors produced a concentration dependent reduction of the plasma crystallization rate in the PD assay by up to 80%. SNF472 was found to be the most potent inhibitor (Figure 1).

Figure 1. Inhibition of calcium phosphate crystallization by calcification inhibitors added in vitro to blank human plasma samples

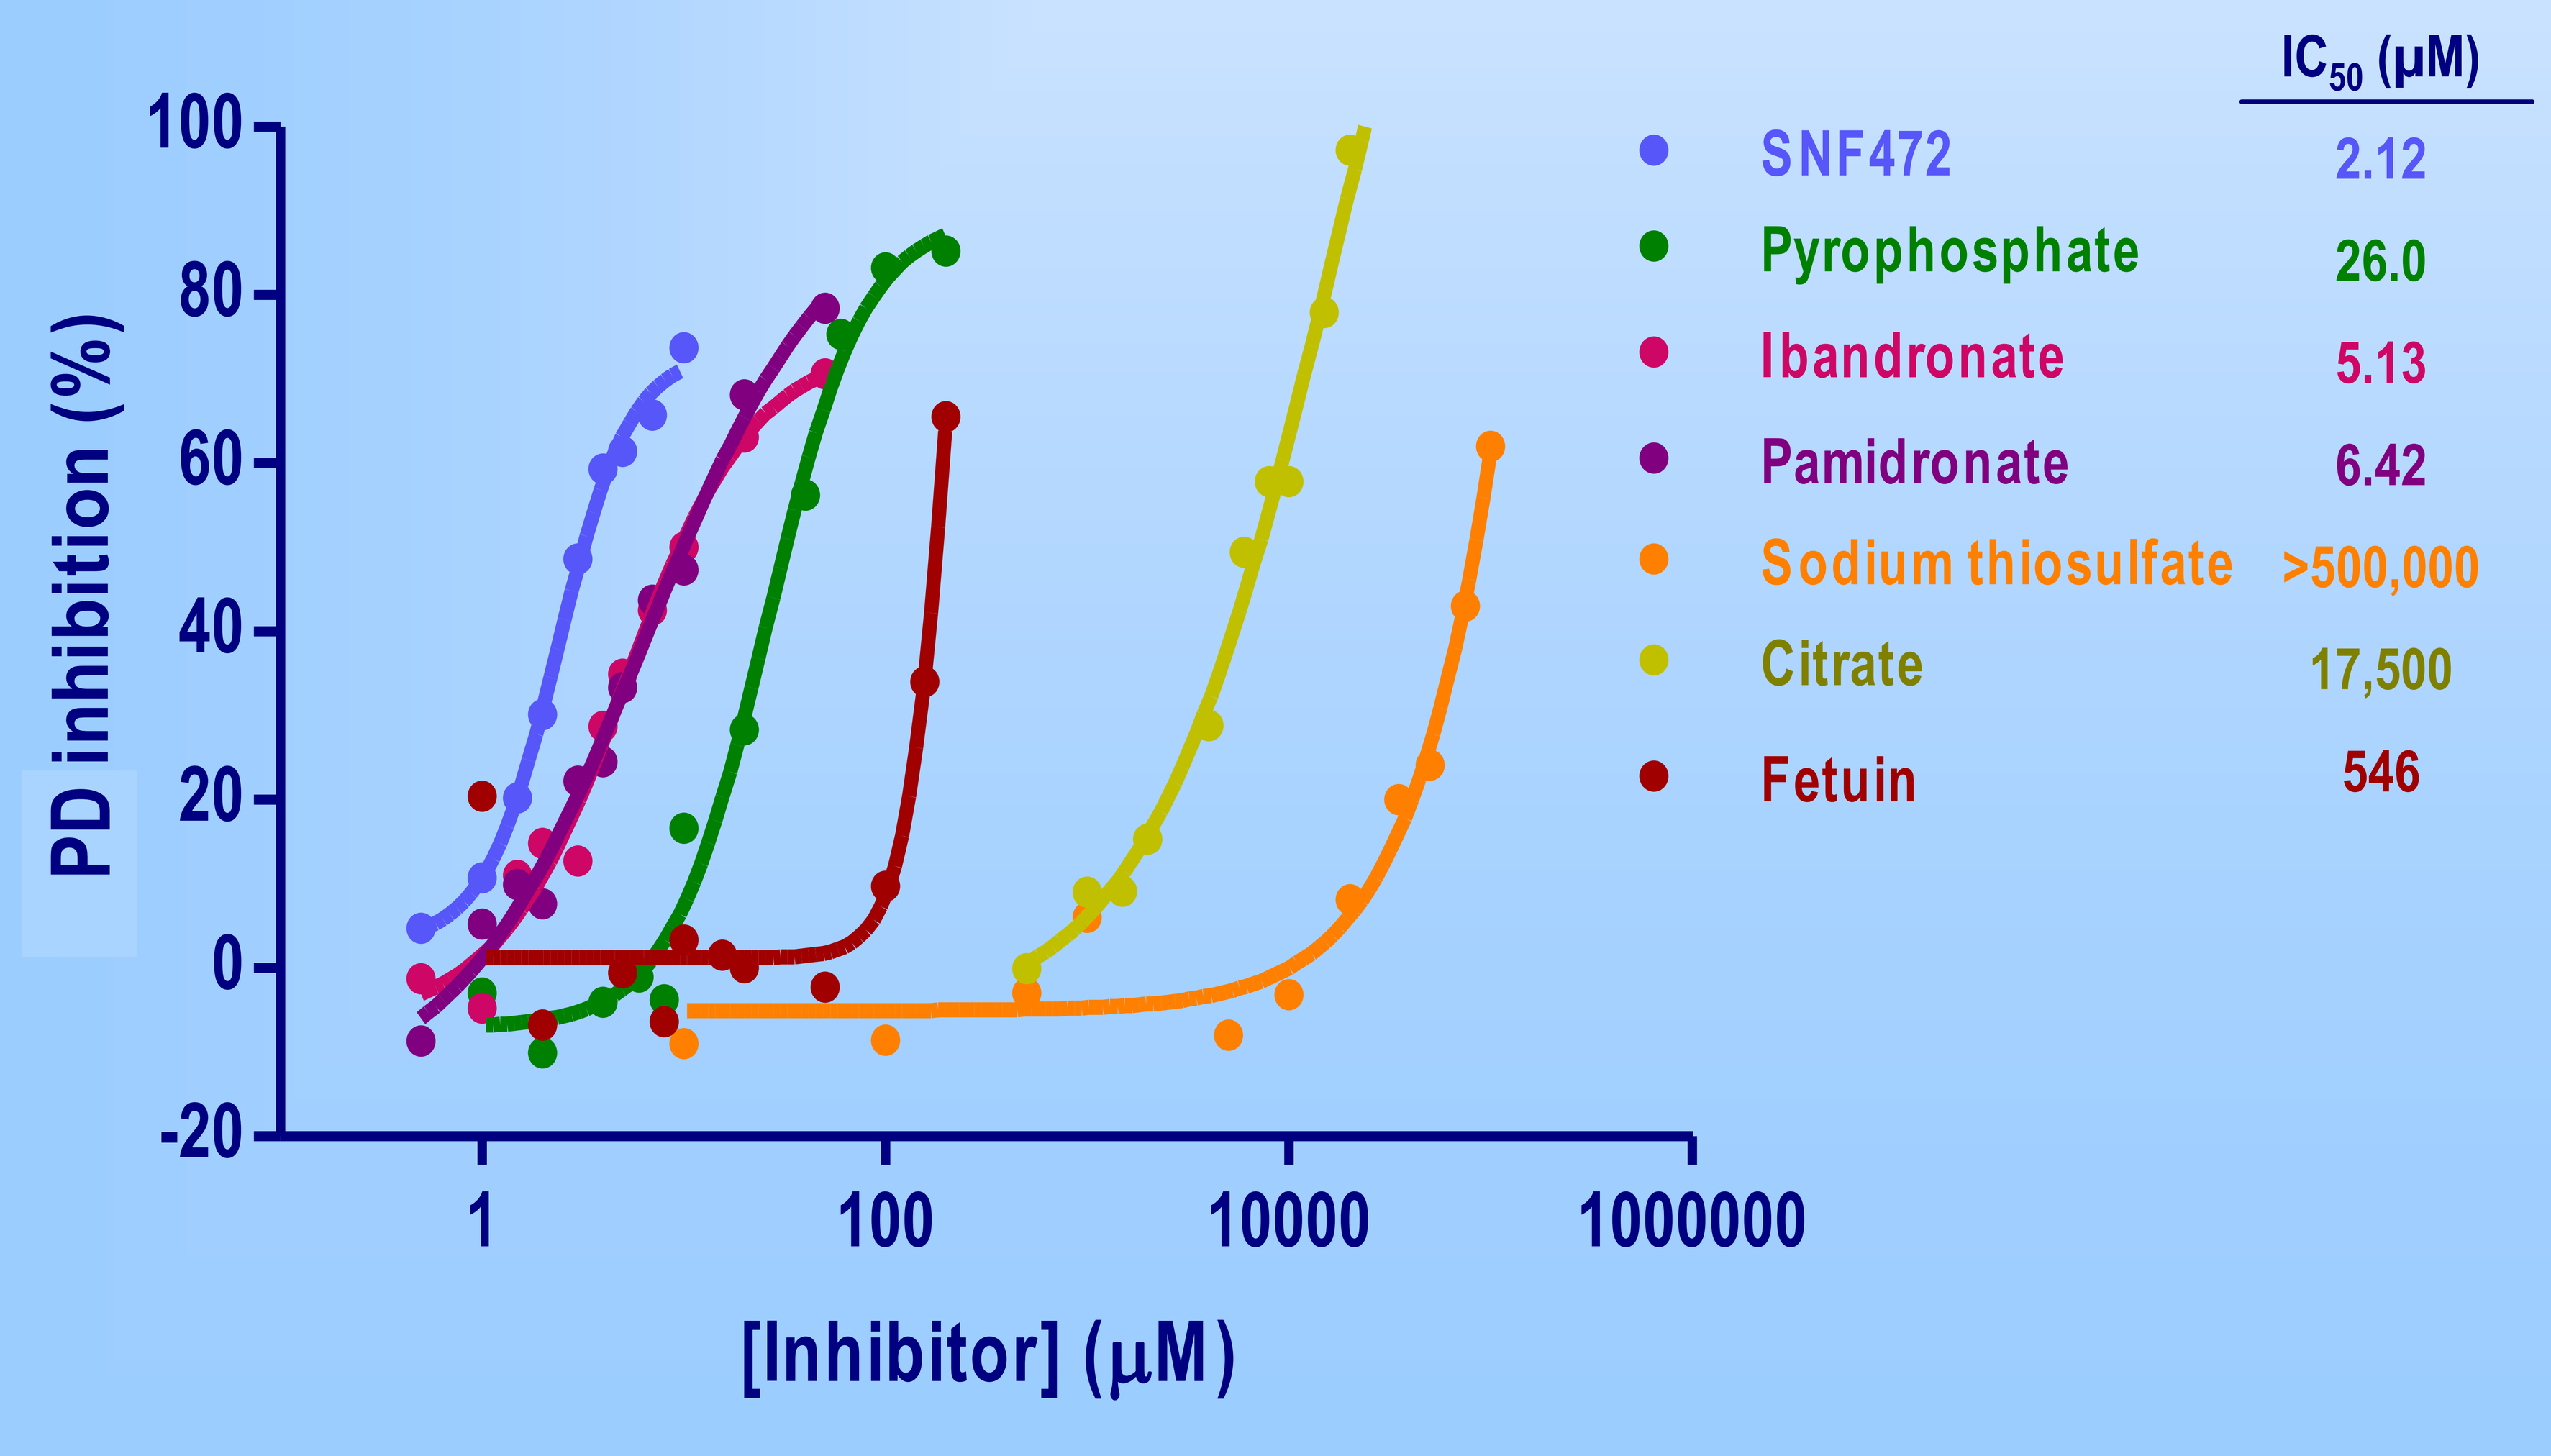
PD inhibition (%)
[Inhibitor] (µM)
1
100
10000
1000000
IC50 (µM)
SNF472 2.12
Pyrophosphate 26.0
Ibandronate 5.13
Pamidronate 6.42
Sodium thiosulfate >500,000
Citrate 17,500
Fetuin 546

Crystallization was induced by the addition of 12.5 mM calcium and 1.5 mM phosphate, and monitored spectrophotometrically in the linear range. Experiments were performed with six replicates per concentration and results represent means.

Preclinical validation: When tested in animals, the in vivo inhibition of CVC (Figures 2A, 2B), in the rat model of calcification, was accompanied by an inhibition of calcium phosphate crystallization in plasma samples obtained at t_{max} (Figure 2C). A direct correlation was observed between the in vivo reduction of CVC and the PD assay results (Figure 2D).

Figure 2. Inhibition of (A) aorta and (B) heart calcification in vivo and (C) plasma ex vivo calcification in rats treated with 4 hours intravenous infusion of SNF472 after calcification induction with 3 x 75 kIU/kg vitamin D. (D) Correlation between in vivo and ex vivo inhibition

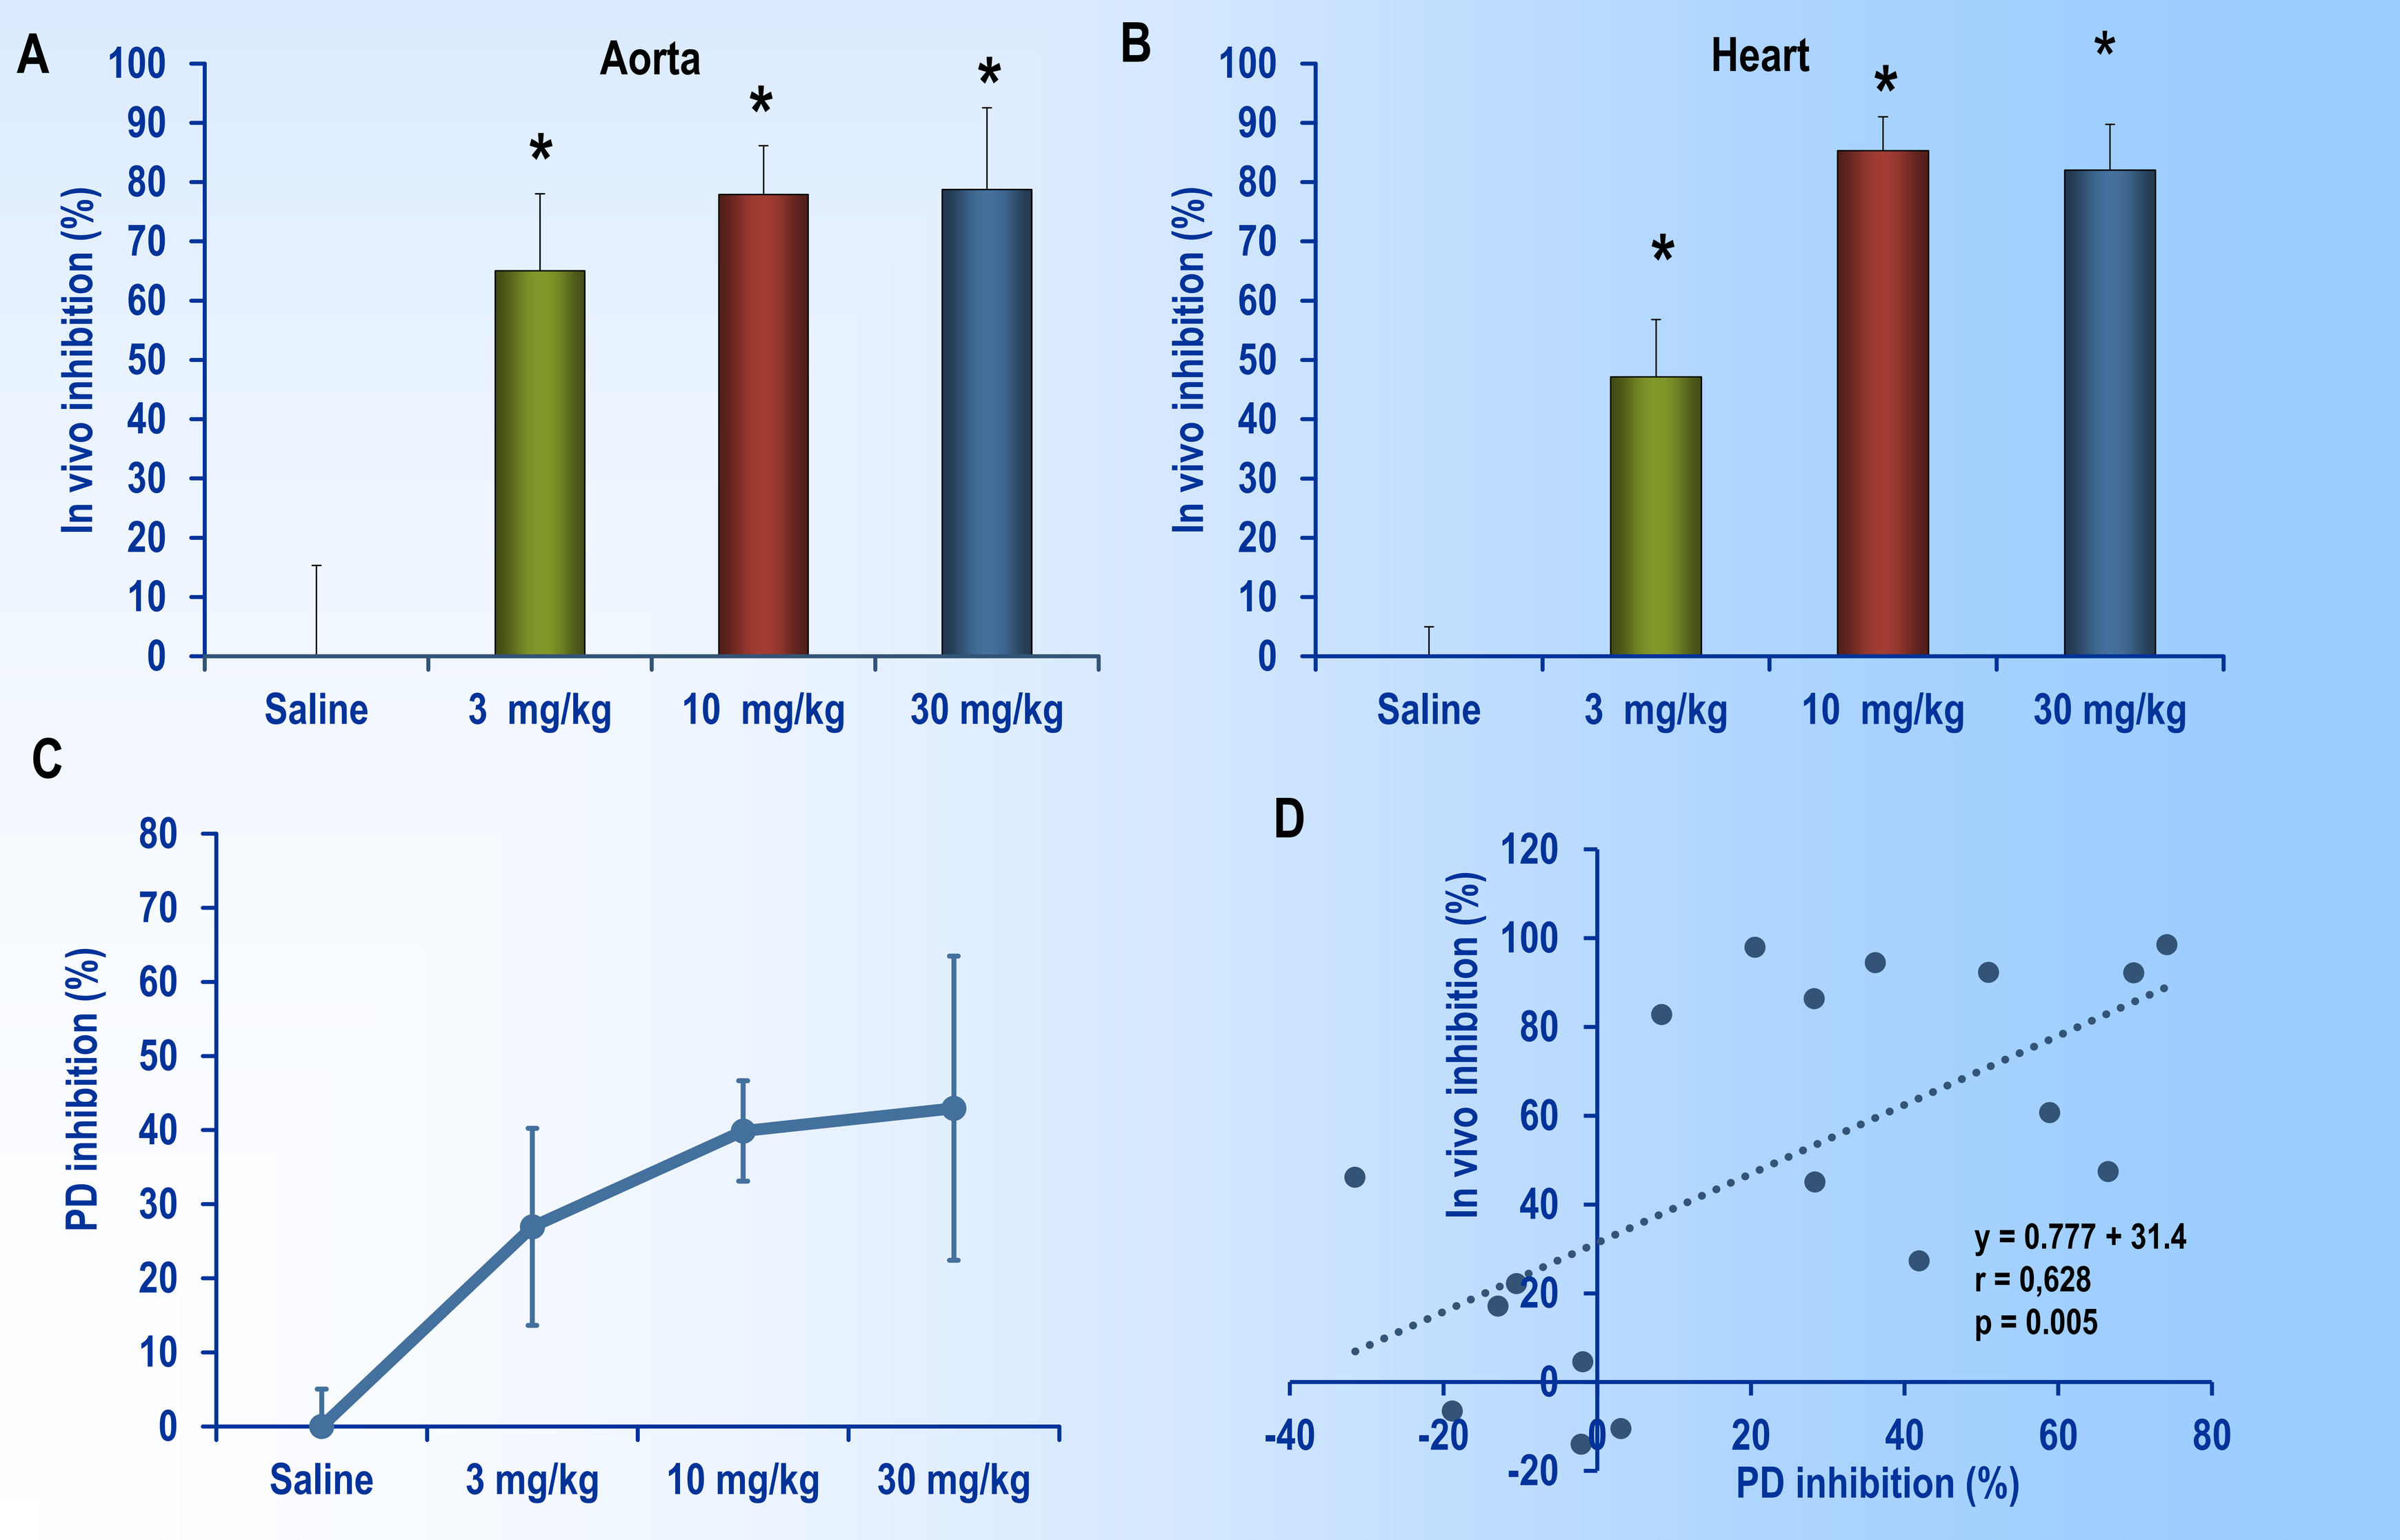
A
Aorta
In vivo inhibition (%)
Saline
3 mg/kg
10 mg/kg
30 mg/kg
B
Heart
In vivo inhibition (%)
C
PD inhibition (%)
D
In vivo inhibition (%)
PD inhibition (%)
y = 0.777 + 31.4
r = 0,628
p = 0.005

(A, B, C) Results represent mean ± S.E.M. One-way ANOVA. (*) Differences vs Saline, $p < 0.05$. (D) Bivariate analysis

Clinical validation: When tested in humans, intravenous infusion of SNF472 in hemodialysis patients at doses from 3 to 20 mg/kg significantly inhibited the induction of HAP crystallization in plasma samples by up to 84% with an IC_{50} of 2.18 mg/kg (Figure 3). No significant differences were observed when SNF472 was dosed between 3 and 20 mg/kg.

Figure 3. Inhibition of induction of hydroxyapatite crystallization in plasma samples after SNF472 4-hour intravenous infusion in HD patients

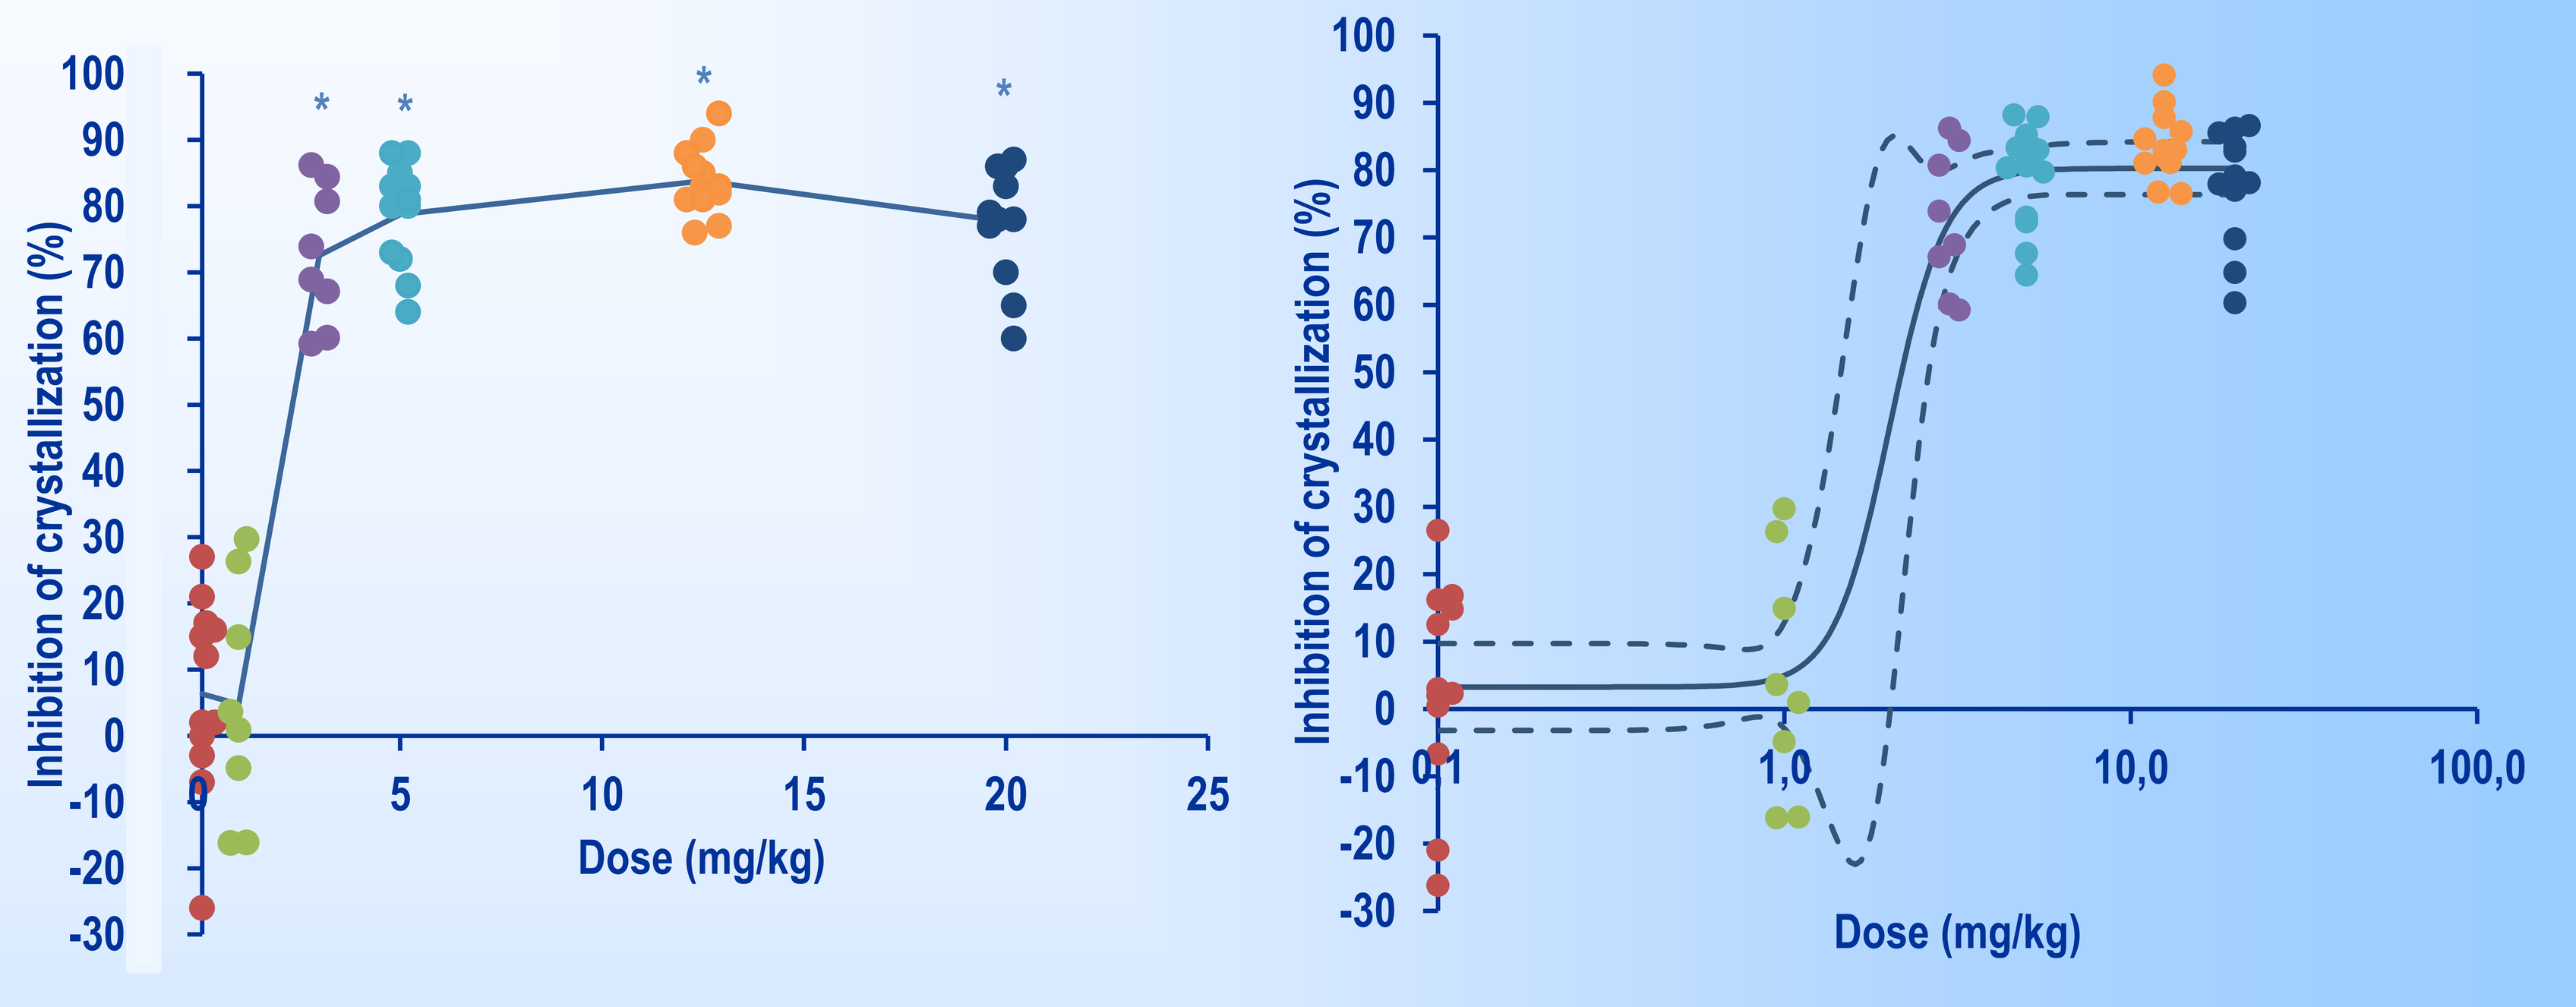
Inhibition of crystallization (%)
Dose (mg/kg)
Inhibition of crystallization (%)
Dose (mg/kg)

(A) Dose-response linear modelling. Solid line connects the means of the doses; (B) Four-parameter dose-response modelling. Solid line represent the dose-response curve calculated from the individual data points. Dashed lines represent the 90% confidence interval of the model. One-way ANOVA: (*) significant differences vs placebo and 1 mg/kg; $p < 0.001$.

CONCLUSIONS

1. A novel PD assay was developed to evaluate and discriminate between inhibitory calcification activity of compounds, such as polyphosphates.

2. These PD measurements predicted CVC inhibition in animal models.

3. This PD assay is proposed as a valuable tool to evaluate the effect of potential CVC inhibitors in clinical trials in hemodialysis patients.

This study was supported by FEDER funds ISCIII RETIC REDINREN RD012/20021 and RETOS COLABORACIÓN: RTC-2014-2460-1 ISCIII grant (Ministerio de Economía y Competitividad. Government of Spain).

GOBIERNO DE ESPAÑA — MINISTERIO DE ECONOMÍA Y COMPETITIVIDAD

Cofinanciado por el Fondo Europeo de Desarrollo Regional (FEDER). Unión Europea. Una manera de hacer Europa

54 ERA
667--MP
Dialysis - Bone disease
Carolina Salcedo
DOI: 10.3252/pso.eu.54ERA.2017
ePosters supported by F. Hoffmann- La Roche Ltd.