MP608

REAL-WORLD OUTCOMES OF HYPERKALEMIA MANAGEMENT WITH PATIROMER IN END-STAGE RENAL DISEASE PATIENTS UNDERGOING HEMODIALYSIS IN THE UNITED STATES

Dinesh K. Chatoth, MD[1], Peter M. Wahl, ScD[2], Viatcheslav Rakov, MD[3], Carly R. Van Zandt, MS[4], Kathryn P. Anastassopoulos, MS[2], Sam Colman, MSc[2], Tyler Knight, MS[2], Nina Oestreicher, PhD[5], Ann Mooney, MSN[4], David M. Spiegel, MD[5]

[1]Fresenius Medical Care North America, Waltham, MA, USA, [2]Covance Market Access Inc., Gaithersburg, MD, USA, [3]Vifor Pharma, Glattbrugg, Switzerland, [4]Frenova Renal Research, Waltham, MA, USA, [5]Relypsa Inc., a Vifor Pharma Company, Redwood City, CA, USA.

INTRODUCTION AND AIMS

- Hyperkalemia (HK) is a life-threatening condition that is common in patients with end-stage renal disease (ESRD) and is associated with adverse clinical consequences.
- Currently, there are a limited number of pharmaceutical products to treat and control HK.
- Sodium polystyrene sulfonate (SPS) was the only available potassium binder in the United States (US) until the US Food and Drug Administration approved patiromer in October 2015 for the treatment of HK.[1]
- The current version of the US Prescribing Information (November 2016)[2] for patiromer reflects the most up-to-date pharmacovigilance information and supports the known safety profile of patiromer.
- Use of SPS in daily practice is limited due to inferior tolerability and reported serious gastrointestinal events.[3-7]
- Clinical trials have demonstrated efficacy of patiromer,[8,9] but little has been reported about patiromer in dialysis settings in clinical practice since it became commercially available in the US.
- The aim of this study was to describe the real-world patient characteristics, management, and outcomes of patiromer for treatment of HK in patients undergoing hemodialysis (HD) for ESRD in a large US ESRD provider network.

METHODS

Study Design

- Retrospective, descriptive cohort study of patients undergoing dialysis at Fresenius Kidney Care (FKC) centers using data from July 1, 2015, through October 31, 2016.
- Patients were included in the study who met the following criteria:
 - Initiated patiromer (first recorded prescription) between October 2015 and July 2016.
 - Prescribed permanent, in-center HD at FKC prior to patiromer initiation.
 - Prescribed HD ≥3 times per week.
 - ≥18 years of age.
 - ≥1 serum potassium (mEq/L; sK) laboratory value recorded in the 91 days prior to patiromer initiation.
 - No report of never filling the patiromer prescription.

Definitions

- The following definitions were used:
 - **Index event:** First recorded patiromer prescription at an FKC dialysis center.
 - **Index date:** The start date on the first recorded patiromer prescription.
 - **Pre-index period**: Up to the 91 days prior to the index date.
 - **Post-index period:** The period of continuous patiromer therapy including and following the index date, where continuous therapy ends at the earliest of the following: patiromer stop date of last patiromer prescription, death date, FKC discharge date, or 90 days after index date. If there is a gap between a patiromer prescription stop date and a following patiromer prescription start date, continuous therapy will end with the prior patiromer prescription stop date.

Analysis

- Patient demographics and clinical characteristics were examined during the pre-index period.
- Serum potassium (mEq/L) and potassium baths (1K) were examined during the pre-index and post-index periods.
- Patiromer treatment patterns were examined during the post-index period:
 - Prescribed initiation dose
 - Prescribed daily dose
 - Dose titrations up and down
 - Prescribed length of therapy
- Pre-index sK was the last recorded value in the pre-index period.
- Post-index sK were the last recorded values in the post-index time periods.
 - Week 1: 1-7 days (day 1 is index date)
 - Week 2: 8-14 days
 - Week 3: 15-21 days
 - Week 4: 22-28 days
 - Month 2: 29-59 days
 - Month 3: 60-91 days
- Potassium baths were measured over the entire pre-index and post-index periods.
- Analyses were conducted for all patients and the following pre-index sK subgroups:
 - sK ≤5.5 mEq/L
 - sK >5.5 mEq/L
 - sK >6.5 mEq/L (subset of >5.5 mEq/L)
- Analyses also examined patients who had a prescription for SPS during the pre-index period (SPS-switcher) and those who did not (SPS-naïve).

RESULTS

Patient Population

- Among 317 patiromer initiators at FKC centers, 268 (84.5%) met the selection criteria **(Figure 1).**
- 89 patients (33.2%) switched from SPS to patiromer; 179 (66.8%) were naïve to SPS.

Figure 1. Selection of Patient Population

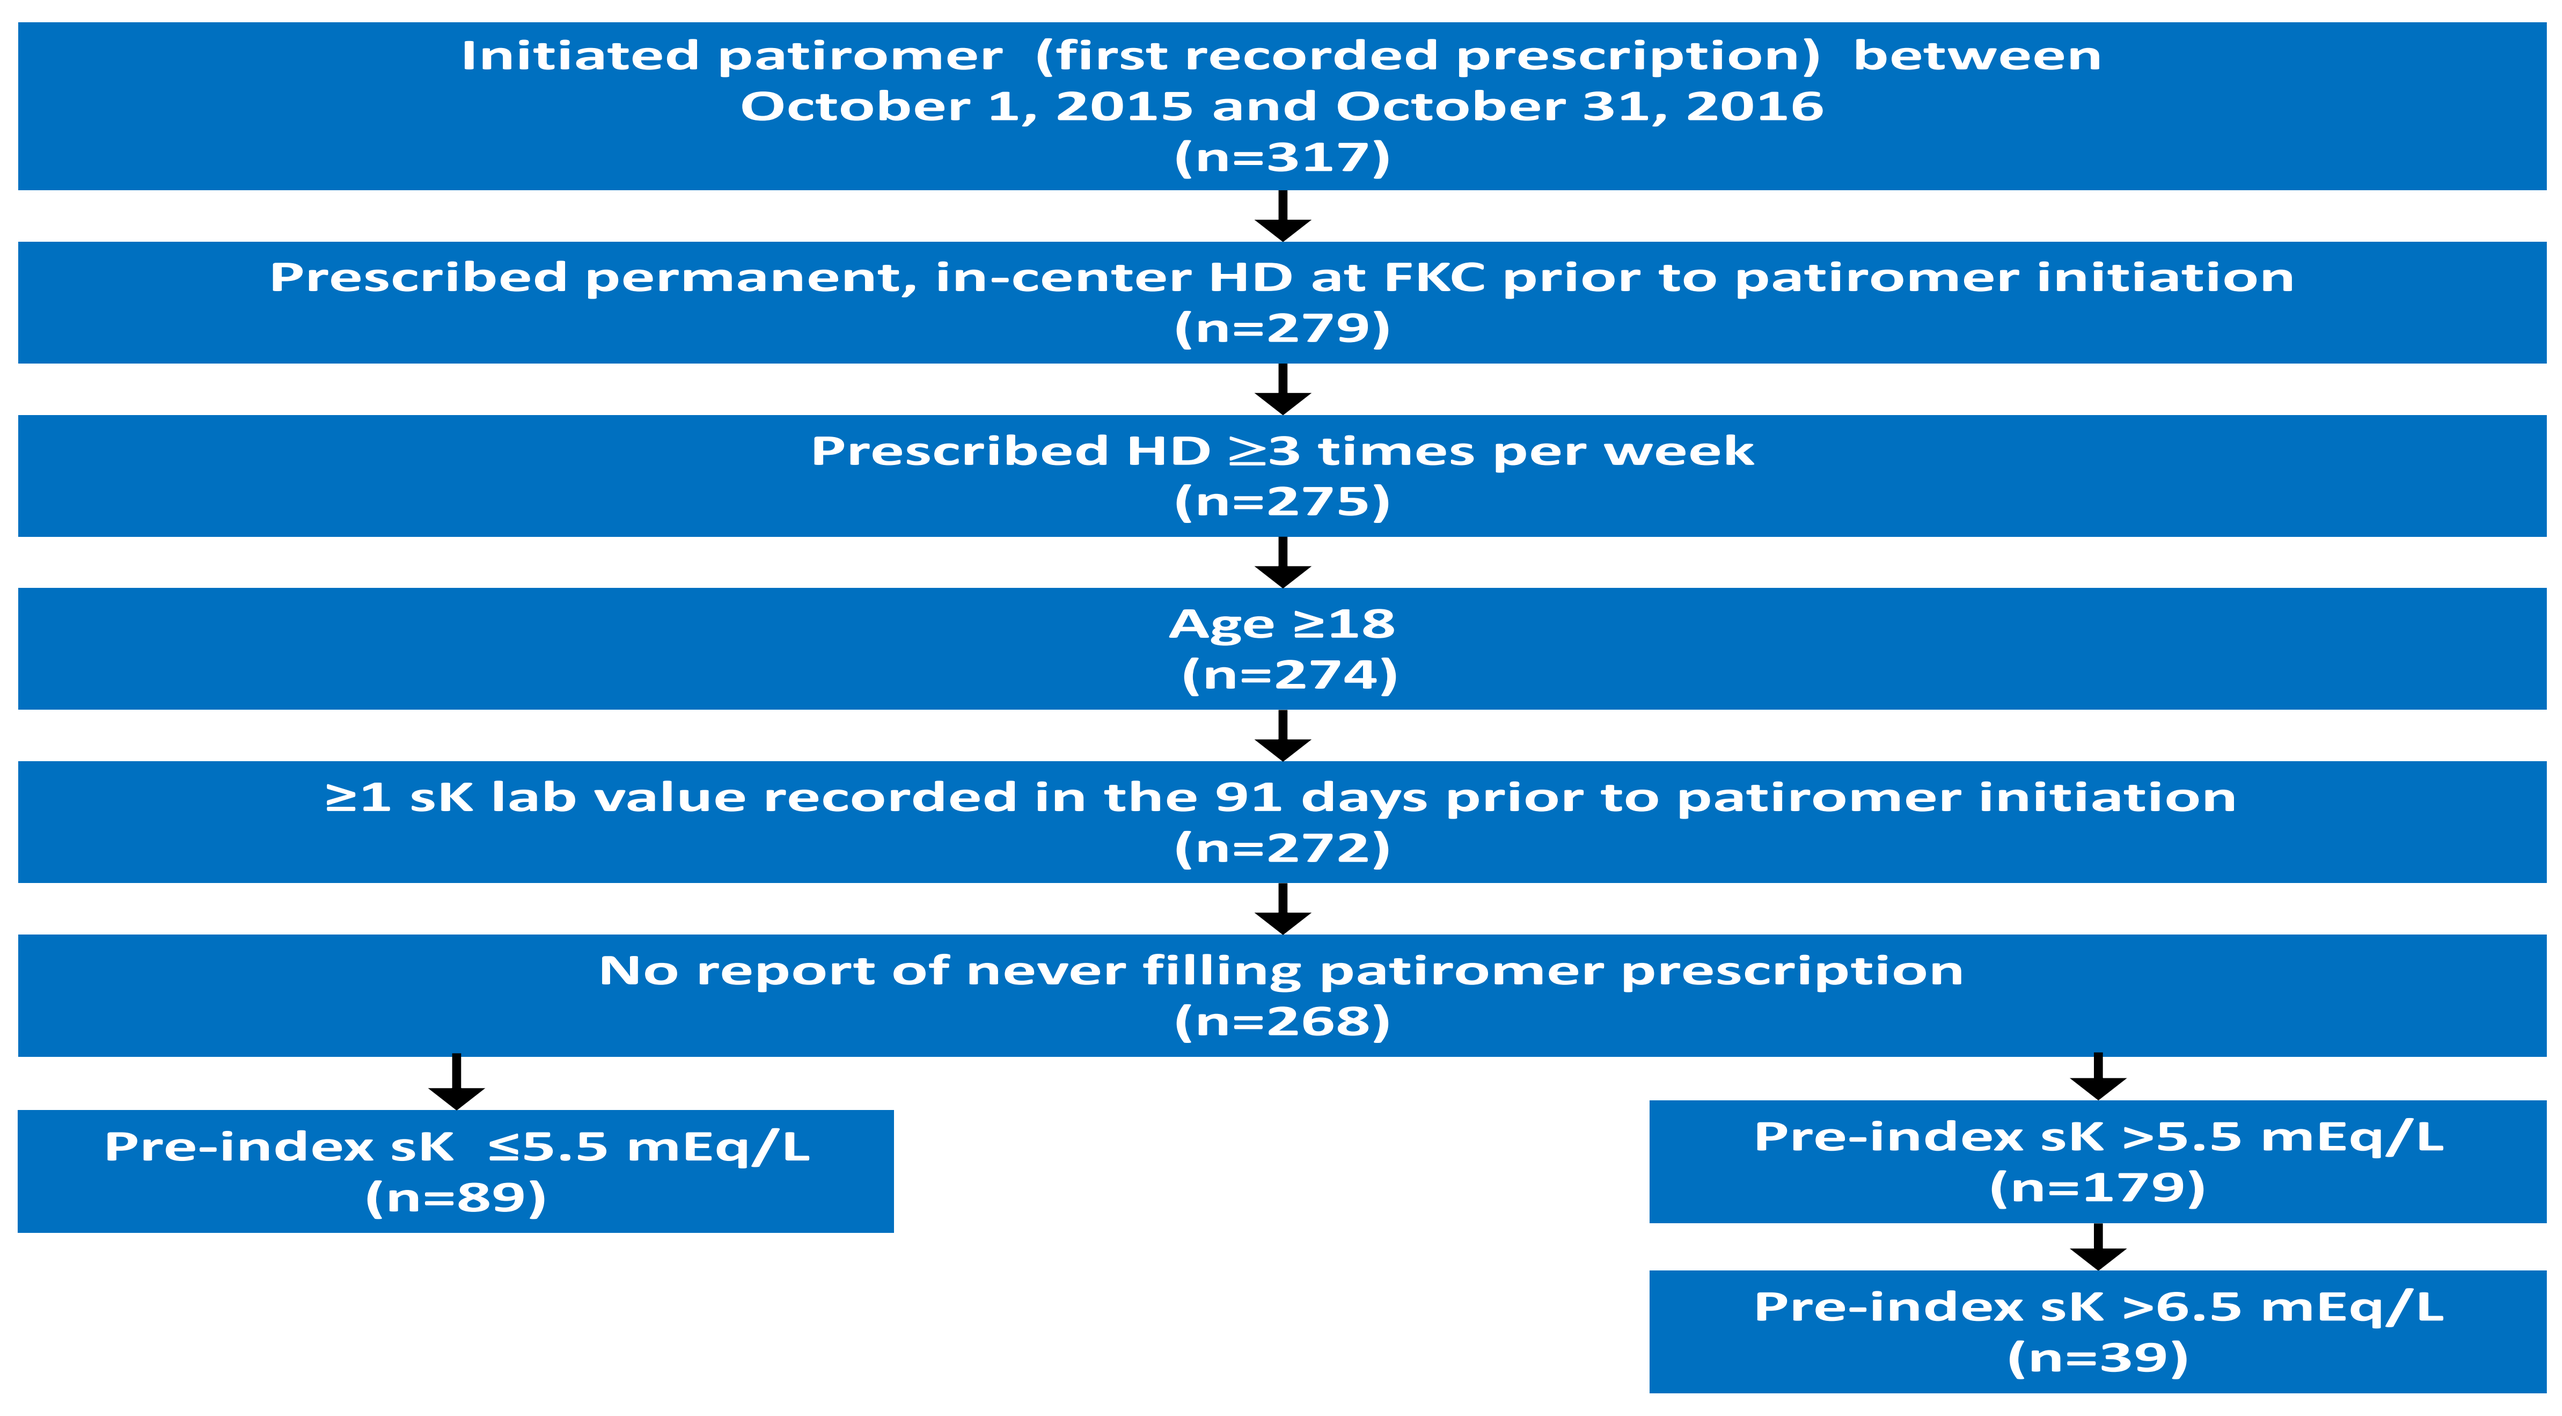
Initiated patiromer (first recorded prescription) between October 1, 2015 and October 31, 2016 (n=317)
Prescribed permanent, in-center HD at FKC prior to patiromer initiation (n=279)
Prescribed HD ≥3 times per week (n=275)
Age ≥18 (n=274)
≥1 sK lab value recorded in the 91 days prior to patiromer initiation (n=272)
No report of never filling patiromer prescription (n=268)
Pre-index sK ≤5.5 mEq/L (n=89)
Pre-index sK >5.5 mEq/L (n=179)
Pre-index sK >6.5 mEq/L (n=39)

FKC, Fresenius Kidney Care; HD, hemodialysis; sK, serum potassium.

Demographics and pre-index clinical characteristics (Table 1)

- Mean age was 57.5 years.
- Over half were male (55.2%).
- Majority (77.2%) of patients were white.
- Average body mass index was 27.4 kg/m².
- Average time on dialysis was 4.9 years.
- The two most common causes of ESRD were type 2 diabetes (41.4%) and hypertension (22.8%).

Table 1. Demographics and Pre-index Clinical Characteristics

Demographic/Clinical Characteristic	N=268
Age (years), mean (SD)	57.5 (13.1)
Male, n (%)	148 (55.2)
Race, n (%)	
White	207 (77.2)
Black	36 (13.4)
Other	25 (9.4)
Body mass index (kg/m²), mean (SD)	27.4 (6.5)
Time on dialysis (years), mean (SD)	4.9 (3.6)
Potassium bath (1K) received, n (%)	
n (%)	85 (31.7)
Mean number of 1K baths (SD) among those who received at least 1	16.4 (12.5)
ESRD cause, n (%), top 5	
Type 2 diabetes mellitus	111 (41.4)
Hypertension	61 (22.8)
Nephritis	19 (7.1)
Type 1 diabetes mellitus	18 (6.7)
Polycystic kidney disease	16 (6.0)

ESRD, end-stage renal disease; SD, standard deviation.

Patiromer treatment patterns

- The majority (71.6%) of patients initiated patiromer therapy on 8.4 g/day **(Figure 2)**.
- Another 21.3% of patients initiated patiromer therapy on <8.4 g/day **(Figure 2)**, resulting in a prescribed initiation average daily dose of 7.8 g/day.
- Few patients were titrated up (7.1%) or down (3.0%).
- Average daily dose during the post-index period was 7.9 g/day.
- 201 patients (75.0%) had at least 91 days of continuous patiromer therapy.

Figure 2. Patiromer Prescribed Initiation Dose

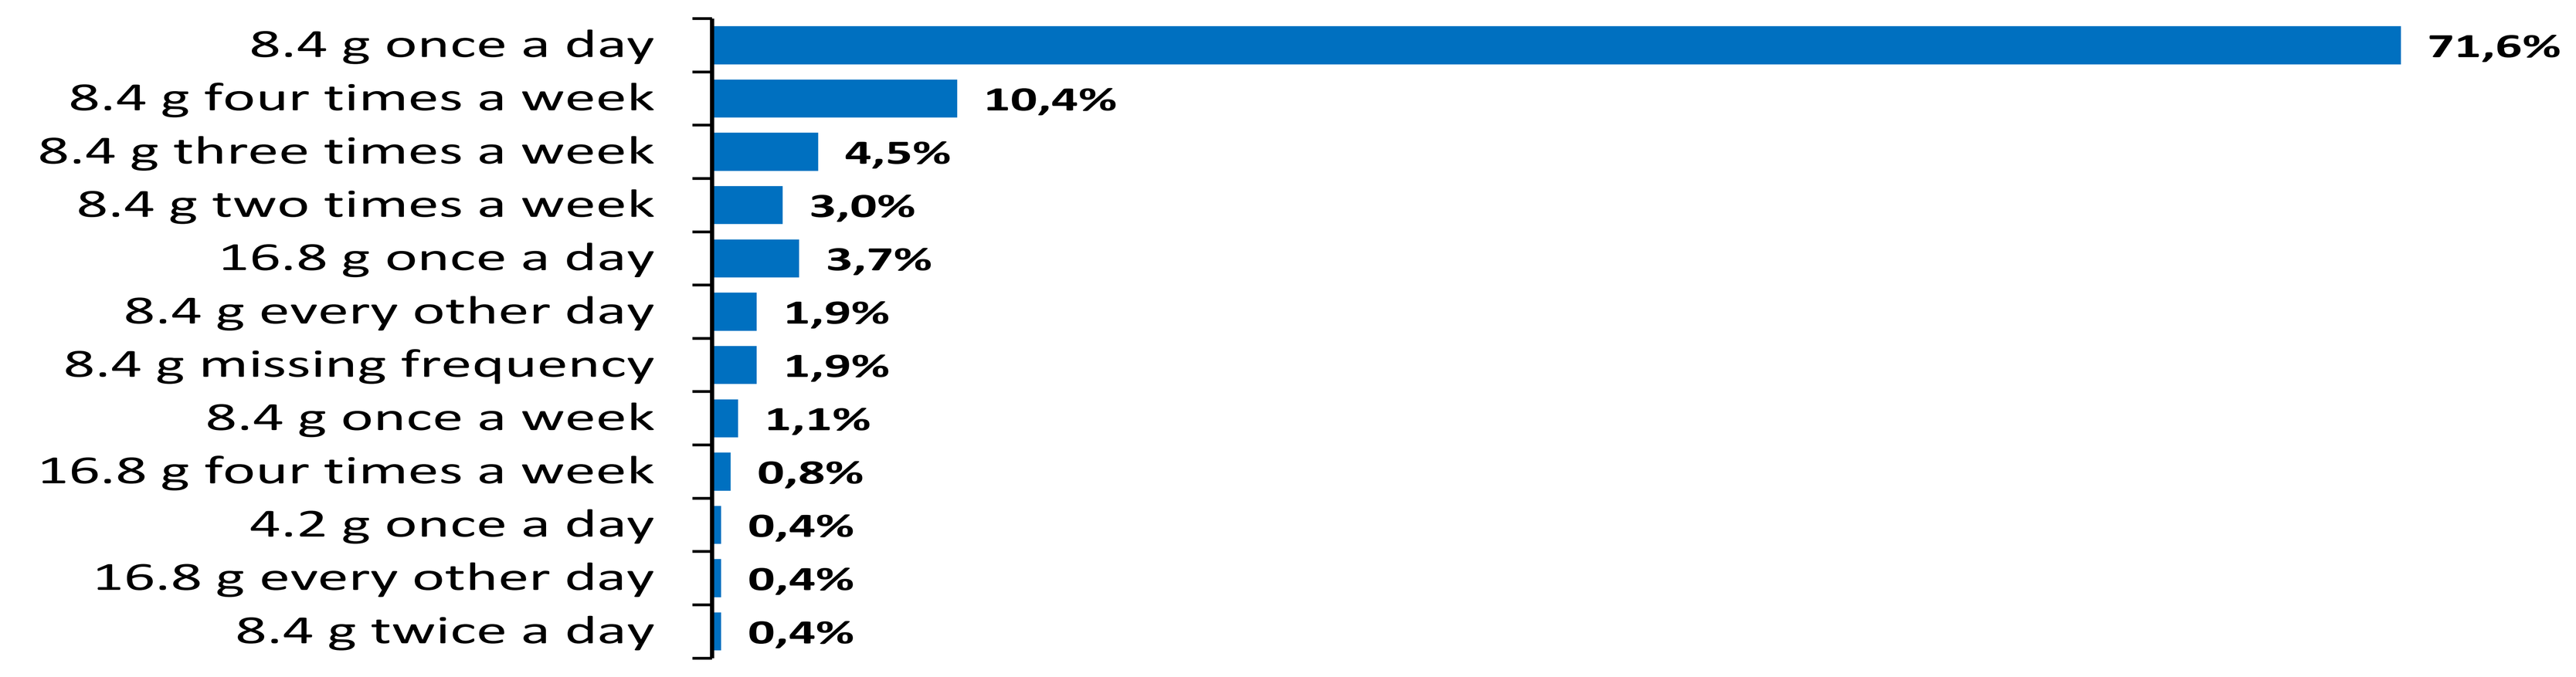
8.4 g once a day 71,6%
8.4 g four times a week 10,4%
8.4 g three times a week 4,5%
8.4 g two times a week 3,0%
16.8 g once a day 3,7%
8.4 g every other day 1,9%
8.4 g missing frequency 1,9%
8.4 g once a week 1,1%
16.8 g four times a week 0,8%
4.2 g once a day 0,4%
16.8 g every other day 0,4%
8.4 g twice a day 0,4%

Serum potassium pre- and post-index (Figure 3)

- Average sK was 5.8 mEq/L in the pre-index period.
- Overall, sK decreased 0.3 mEq/L by week 1 and remained decreased by 0.4 mEq/L at month 2.
- Within the subgroups, sK decreased most among those with higher pre-index sK:
 - ≤5.5 mEq/L subgroup: no decrease
 - >5.5 mEq/L subgroup: 0.6 mEq/L by week 1
 - >6.5 mEq/L subgroup: 1.3 mEq/L by week 1
- Results were similar for SPS-switcher and SPS-naïve patients.
 - SPS-switcher: sK decreased 0.3 mEq/L by week 1
 - SPS-naïve: sK decreased 0.4 mEq/L by week 1

Figure 3. Mean Serum Potassium (mEq/L) Before and After Patiromer Initiation

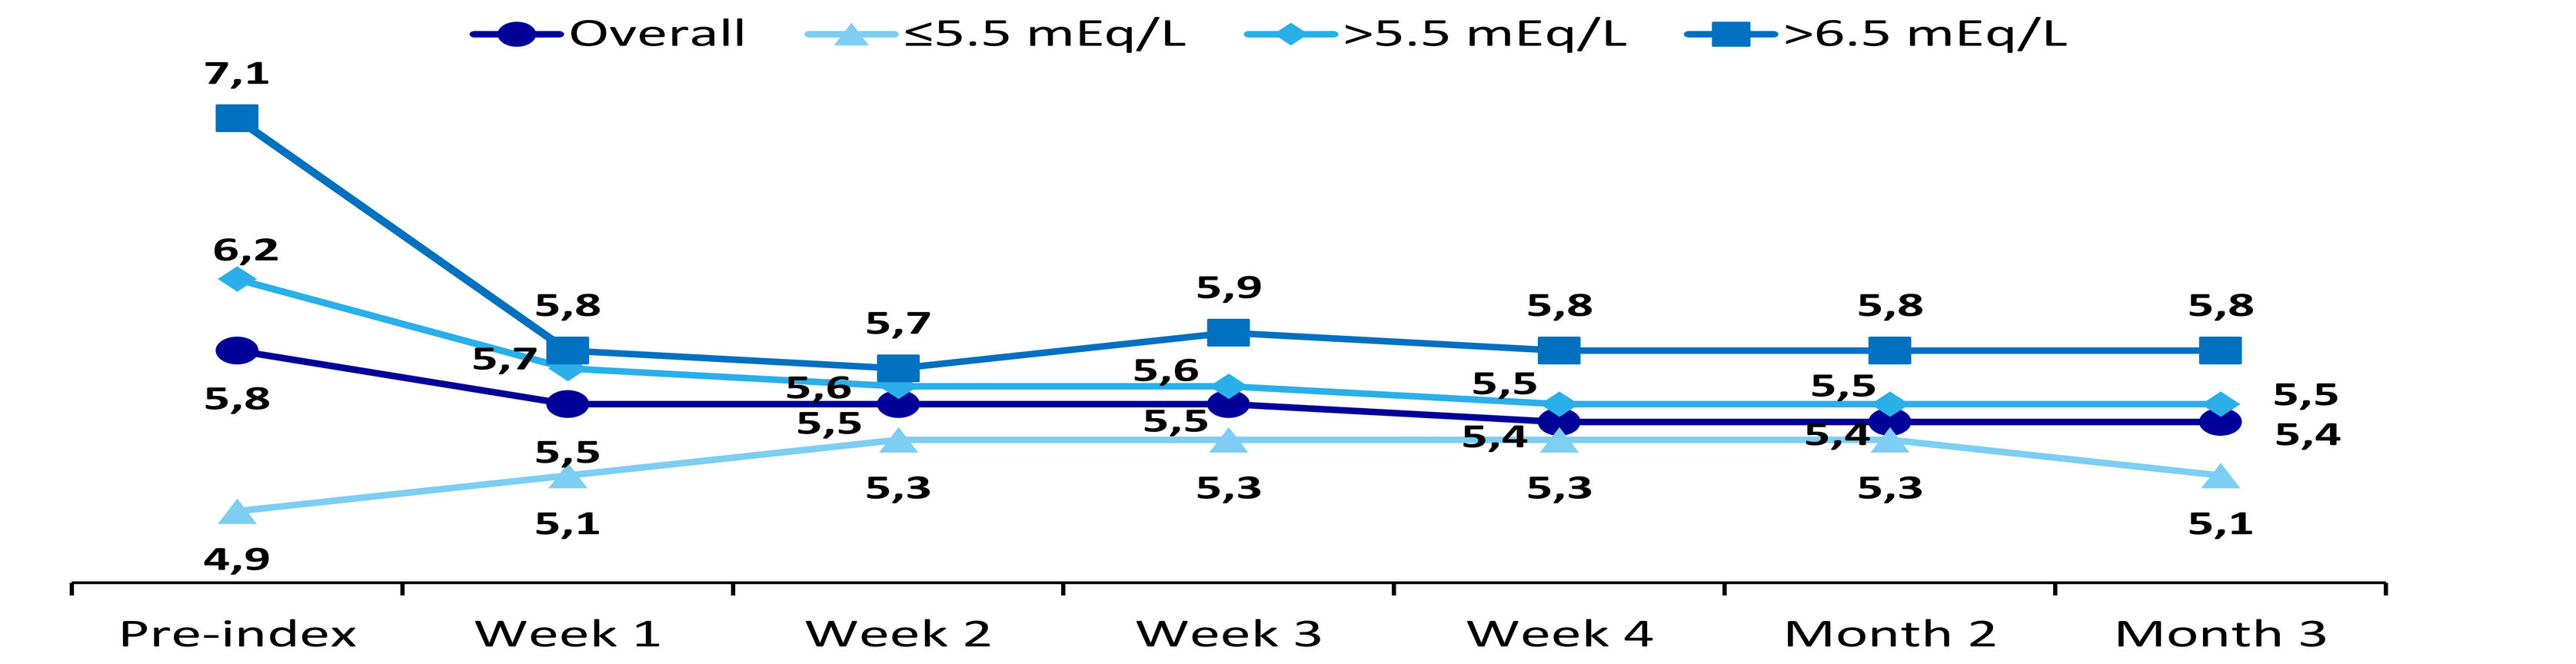
Overall
≤5.5 mEq/L
>5.5 mEq/L
>6.5 mEq/L
7,1
6,2
5,8
4,9
5,8
5,7
5,5
5,1
5,7
5,6
5,5
5,3
5,9
5,6
5,5
5,3
5,8
5,5
5,4
5,3
5,8
5,5
5,4
5,3
5,8
5,5
5,4
5,1
Pre-index
Week 1
Week 2
Week 3
Week 4
Month 2
Month 3

Potassium baths (1K) pre- and post-index

- The number of patients treated with at least one 1K bath decreased.
 - Overall: 4.8% (31.7% to 26.9%) **(Figure 4)**
 - ≤5.5 mEq/L subgroup: 10.1% (29.2% to 19.1%)
 - >5.5 mEq/L subgroup: 2.3% (33.0% to 30.7%)
 - >6.5 mEq/L subgroup: 2.6% (46.2% to 43.6%)
 - SPS-switcher patients: 3.4% (23.6% to 20.2%)
 - SPS-naïve patients: 5.6% (35.8% to 30.2%)
- Among patients who received 1K baths, the mean number of treatments using a 1K bath decreased.
 - Overall: 1.7 (16.4 to 14.7) **(Figure 4)**
 - ≤5.5 mEq/L subgroup: 4.4 (17.5 to 13.1)
 - >5.5 mEq/L subgroup : 0.7 (15.9 to 15.2)
 - >6.5 mEq/L subgroup : 2.1 (15.7 to 13.6)
 - SPS-switcher patients: 2.8 (18.4 to 15.6)
 - SPS-naïve patients: 1.4 (15.8 to 14.4)

Figure 4. Potassium Bath (1K) Utilization Before and After Patiromer Initiation

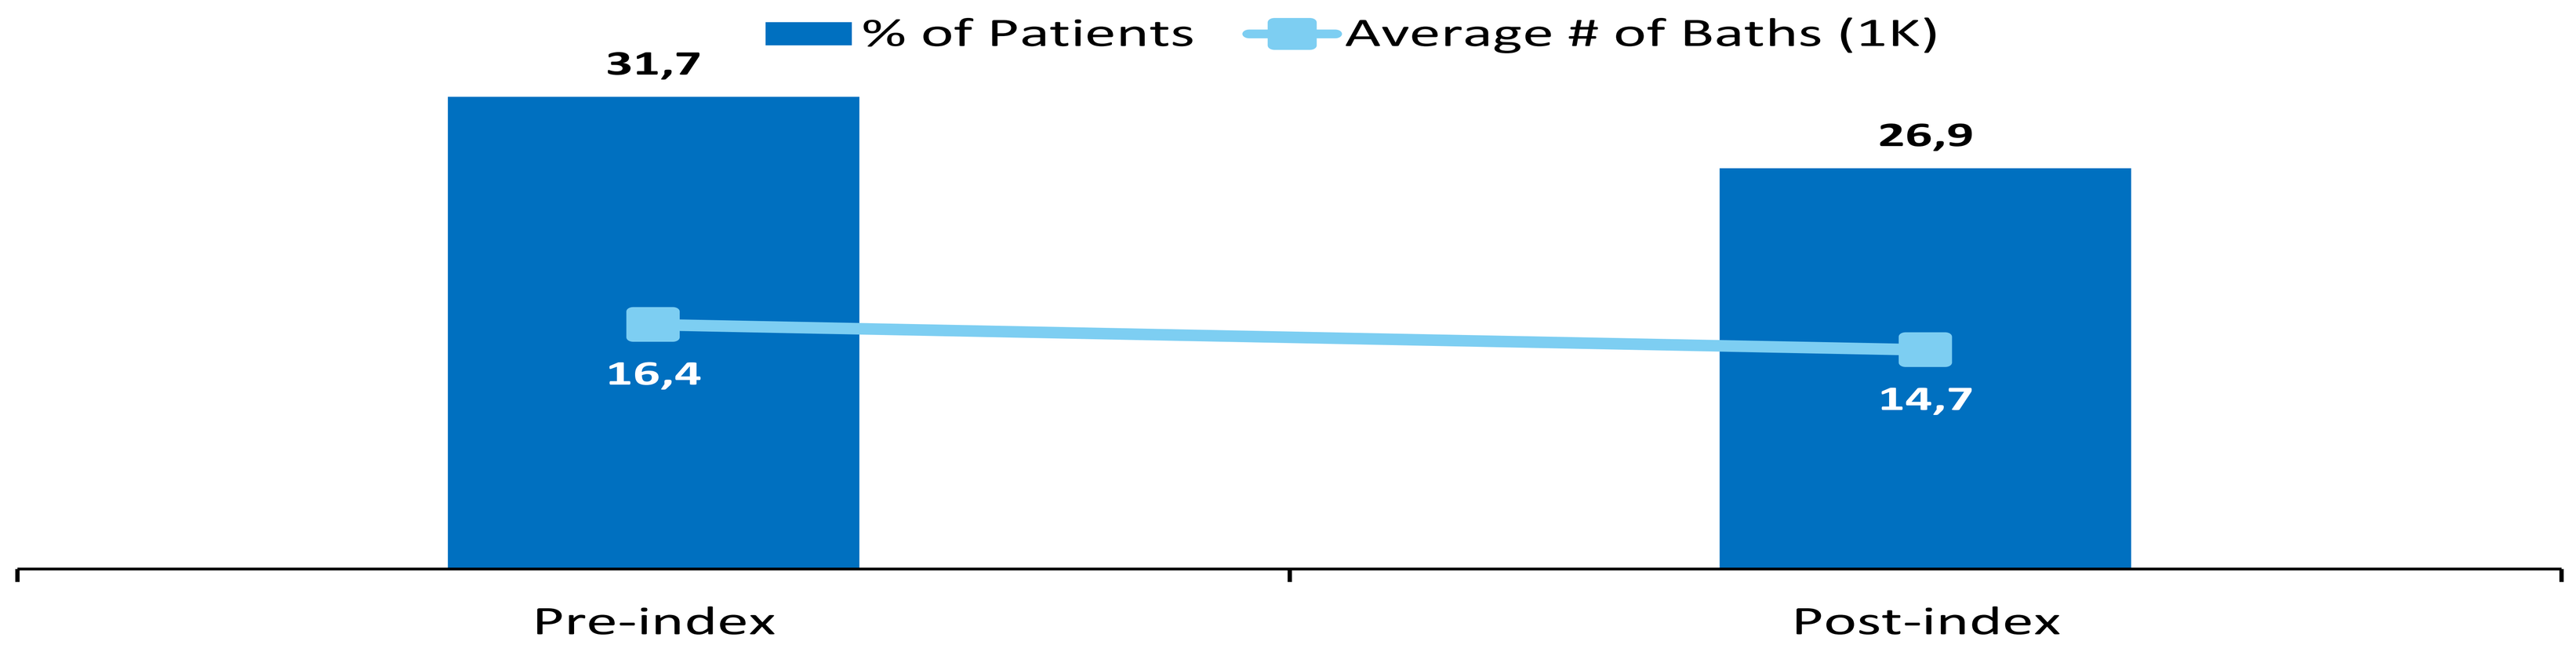
% of Patients
Average # of Baths (1K)
31,7
16,4
26,9
14,7
Pre-index
Post-index

CONCLUSIONS

- These results from observational data on early adopters of patiromer in FKC dialysis centers in the US demonstrate that hyperkalemia management with patiromer lowers serum potassium in ESRD patients undergoing hemodialysis.
- The greatest effect was observed among patients with severe hyperkalemia (pre-index sK >6.5 mEq/L) whose average daily patiromer dose was similar to that of less severe patients.
- The occurrence and frequency of potassium baths (1K) decreased in this population after initiating patiromer for hyperkalemia treatment.
- Results for SPS-naïve and SPS-switcher patients were similar for serum potassium but differences were observed in the need for 1K baths.
- The predominant use of the recommended starting dose of 8.4 g/day as maintenance in this population supports the feasibility of this dose for long-term serum potassium control.

References

1. US Department of Health and Human Services. US Food and Drug Administration. http://www.fda.gov/NewsEvents/Newsroom/PressAnnouncements/ucm468546.htm. Updated October 22, 2015. Accessed November 10, 2016. 2. Relypsa Inc. (2016). Retrieved from https://www.accessdata.fda.gov/drugsatfda_docs/label/2016/205739s009lbl.pdf. Accessed April 24, 2017. 3. Palmer BF. *N Engl J Med.* 2004;351(6):585-592. 4. Harel Z, et al. *Am J Med.* 2013;126(3):264.e9-264.e24. 5. Nguyen T, et al. *JAAPA*. 2015;28(3):41-45. 6. Hagan AE, et al. *Clin Nephrol*. 2016;85(1):38-43. 7. Montaperto AG, et al. *Curr Med Res Opin*. 2016;32(1):155-164. 8. Bakris GL, et al; AMETHYST-DN Investigators. *JAMA*. 2015;314(2):151-161. 9. Weir MR, et al; OPAL-HK Investigators. *N Engl J Med*. 2015;372(3):211-221.

Presented at the 54th ERA–EDTA Congress, 3–6 June, 2017, Madrid, Spain.

Supported by Relypsa, Inc., a Vifor Pharma Company

54 ERA 608--MP

Dialysis - Cardiovascular complications II

Dinesh Chatoth

DOI: 10.3252/pso.eu.54ERA.2017

ePosters supported by F. Hoffmann- La Roche Ltd.

PosterSessionOnline.com