계명대학교 동산의료원
Keimyung University Dongsan Medical Center

Clinical Outcomes According to the Types of Vascular Access in the Very Elderly Hemodialysis Patients: Single Center Experience

Woo Yeong Park[1,3], Seong Sik Kang[1,3], Yaerim Kim[1,3], Kyubok Jin[1,3], Sung Bae Park[1,3], Hyung Tae Kim[1,3], Won Hyun Cho[1,3], Seungyeup Han[1,3]

Department of Internal Medicine[1] and Surgery[2], Keimyung University School of Medicine, Kidney Institute[3], Daegu, Korea

BACKGROUND

The number of elderly patients in need of dialysis is steadily increasing in Korea. The type of vascular access for elderly patients is very important to patient survival of hemodialysis patients. Several studies showed that the prognosis of hemodialysis patients with arteriovenous fistula (AVF) or arteriovenous graft (AVG) were better than that of patients with central venous catheter (CVC) in the non-elderly patients, but may not be insignificant in the very elderly (>75 years old) patients.

AIMS

The aim of this study was to evaluate the clinical outcomes according to the types of vascular access in these patients.

METHODS

♦ Retrospective analysis
♦ Review of medical records
♦ Period: JAN.1. 2005. ~ DEC. 31. 2014.
♦ Patients
- age ≥ 75 years old
- no previous renal replacement therapy
- Survival of initial 3 months of dialysis

RESULTS

The mean age of the patients was 77.7 ± 2.8 years. Male patients were 31 (51.7%). The primary cause of end-stage renal disease included diabetes mellitus 20 (33.3%), hypertension 23 (38.3%), glomerulonephritis 9 (15.0%) and others 8 (13.3%). Forty three (71.7%) patients were initiated dialysis with CVC, included tunneled cuffed catheter 32 (74.4%), before changed to AVF or AVG during the follow-up period. The types of vascular access were composed of AVF 52 (86.7%) and AVG 8 (13.3%). According to the site of AVF, the rate of brachio-cephalic fistula 29 (55.8%) was significantly higher than that of radio-cephalic fistula 23 (44.2%) (p=0.019). However, age, sex and diabetes mellitus were not significantly different between two groups. During the mean follow-up period of 25.2 months, 14 (23.3%) patients underwent secondary AVF or AVG operation, and 19 (31.7%) received percutaneous transluminal angioplasty. Of the 10 (16.7%) patients who died during the follow-up period, 6 died of infection and 4, cardiovascular disease. Kaplan-Meier analysis showed that patient survival rate and primary patency rate with AVF were significantly higher than those of patients with AVG (p=0.040, p=0.013), but secondary patency rate was no significant difference between two groups.

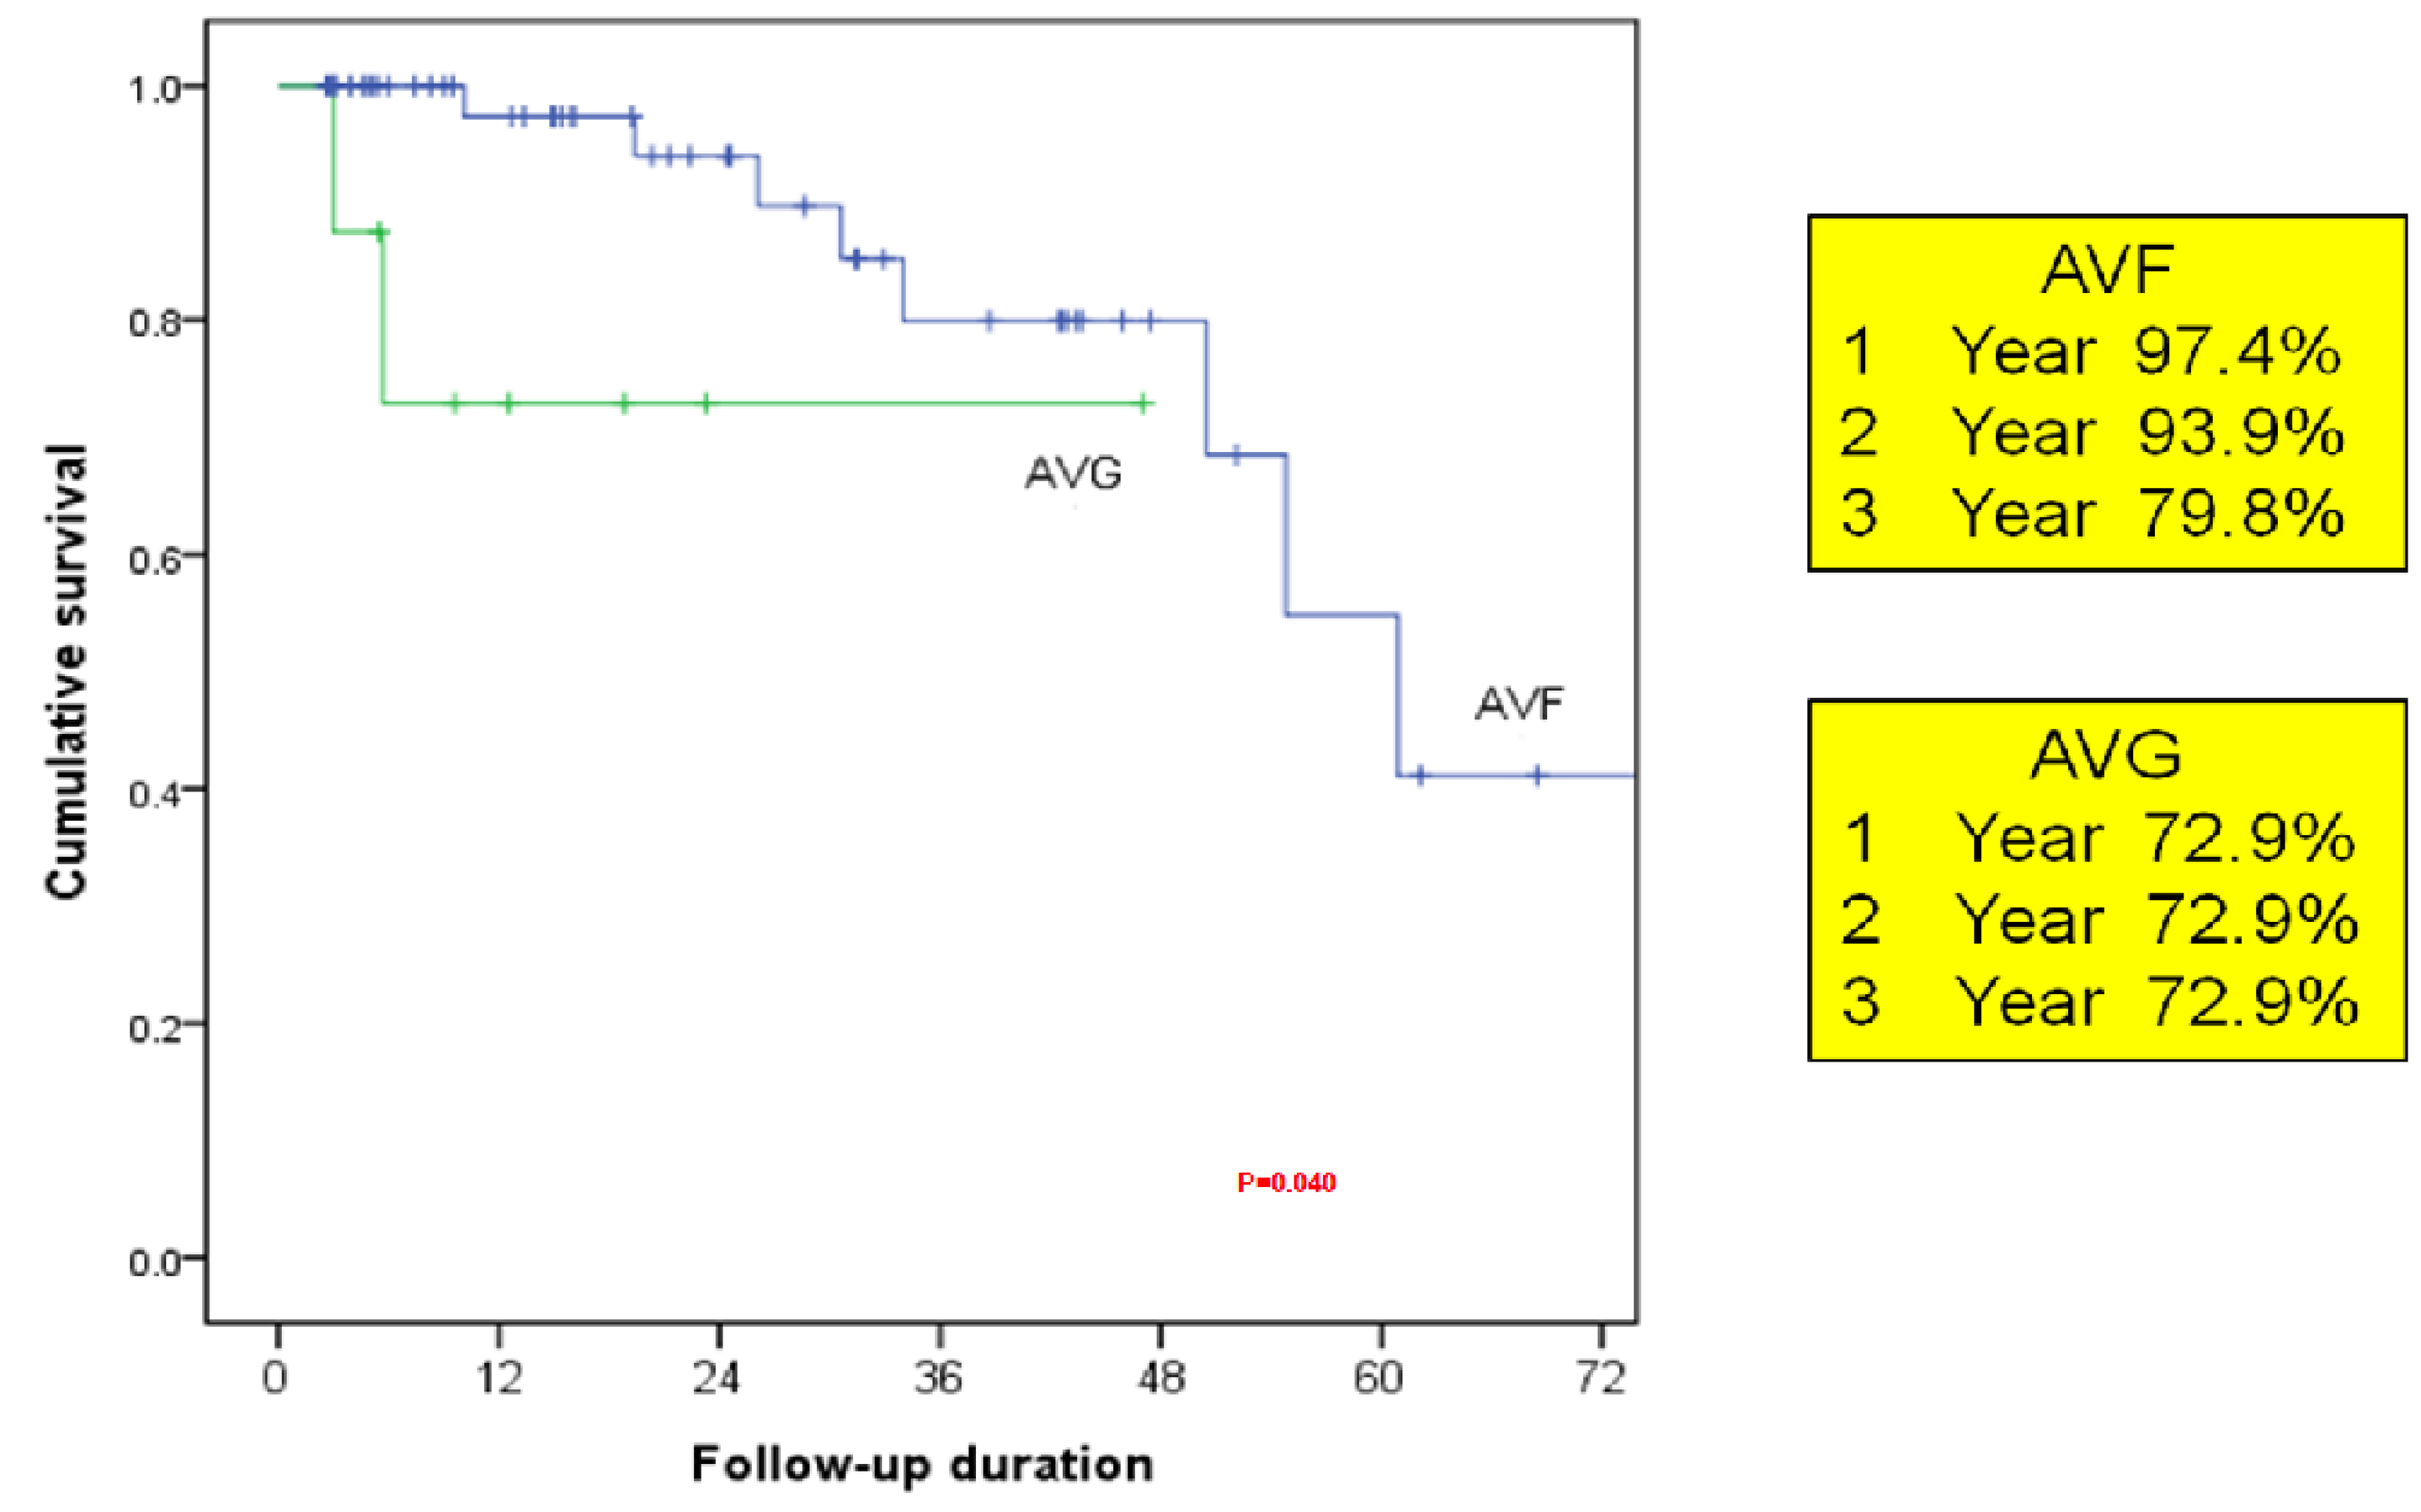
1.0
0.8
0.6
0.4
0.2
0.0
Cumulative survival
0
12
24
36
48
60
72
Follow-up duration
AVG
AVF
P=0.040
AVF
1 Year 97.4%
2 Year 93.9%
3 Year 79.8%
AVG
1 Year 72.9%
2 Year 72.9%
3 Year 72.9%

Figure 1. Patient Survivial according to AVF and AVG

Table 1. Baseline demographic characteristics of the study population (n=60).

Variables	
Age (years)	77.7 ± 2.8
Male, n (%)	31 (51.7)
Dialysis duration, months	25.2 ± 20.9
Cause of end-stage renal disease, n (%)	
Glomerulonephritis	9 (15.0)
Diabetes mellitus	20 (33.3)
Hypertension	23 (38.3)
Others	8 (13.3)
Comorbidity, n (%)	
Diabetes mellitus	24 (40.0)
Hypertension	31 (51.7)
Cardiovascular disease	18 (30.0)
Malignancy	10 (16.7)
Types of vascular access, n (%)	
AVF: AVG	52 (86.7) : 8 (13.3)
Initial vascular access, n (%)	
Central venous catheter	43 (71.7)
Tunneled cuffed catheter	32 (74.4)
Internal jugular catheter	11 (25.6)
Site of vascular access, n (%)	14 (6.8)
Radio-cephalic fistula	23 (44.2)
Brachio-cephalic fistula	29 (55.8)

Table 2. Comparison of clinical and laboratory parameters according to the type of vascular access.

Valuables	AVF (n=52)	AVG (n=8)	*P*-value
Age (year)	77.5±2.6	79.1±4.2	0.146
Male, n (%)	28 (53.8))	3 (37.5)	0.465
Dialysis duration, months	22.5±3.0	22.7±3.3	0.722
Cause of ESRD, n (%)			0.110
Glomerulonephritis	7 (13.5)	2 (25.0)	
Diabetes mellitus	15 (28.8)	5 (62.5)	
Hypertension	22 (42.3)	1 (12.5)	
Others	8 (15.4)	0 (0)	
Comorbidity, n (%)			
Diabetes mellitus	19 (36.5)	5 (62.5)	0.247
Hypertension	28 (53.8)	3 (37.5)	0.465
Cardiovascular disease	16 (30.8)	2 (25.0)	1.000
Malignancy	10 (19.2)	0 (0)	0.330
1st patency period	20.0±16.4	8.2±9.5	0.011
2nd patency period	18.3±16.0	11.8±8.2	0.094

Table 3. Cause of deaths of the study population

Variables	AVF	AVG
Cardiovascular disease	3 (37.5)	1 (50.0)
Infection	5 (62.5)	1 (50.0)

Table 4. Cox regression analysis for patient survivial

Variables	Univariable		Multivariable	
	Exp (β)	*P*-value	Exp (β)	*P*-value
Age	1.163	0.134		
Sex (M)	0.836	0.877		
AVG (vs AVF)	4.910	0.063	0.249	0.019
DM	2.720	0.139		

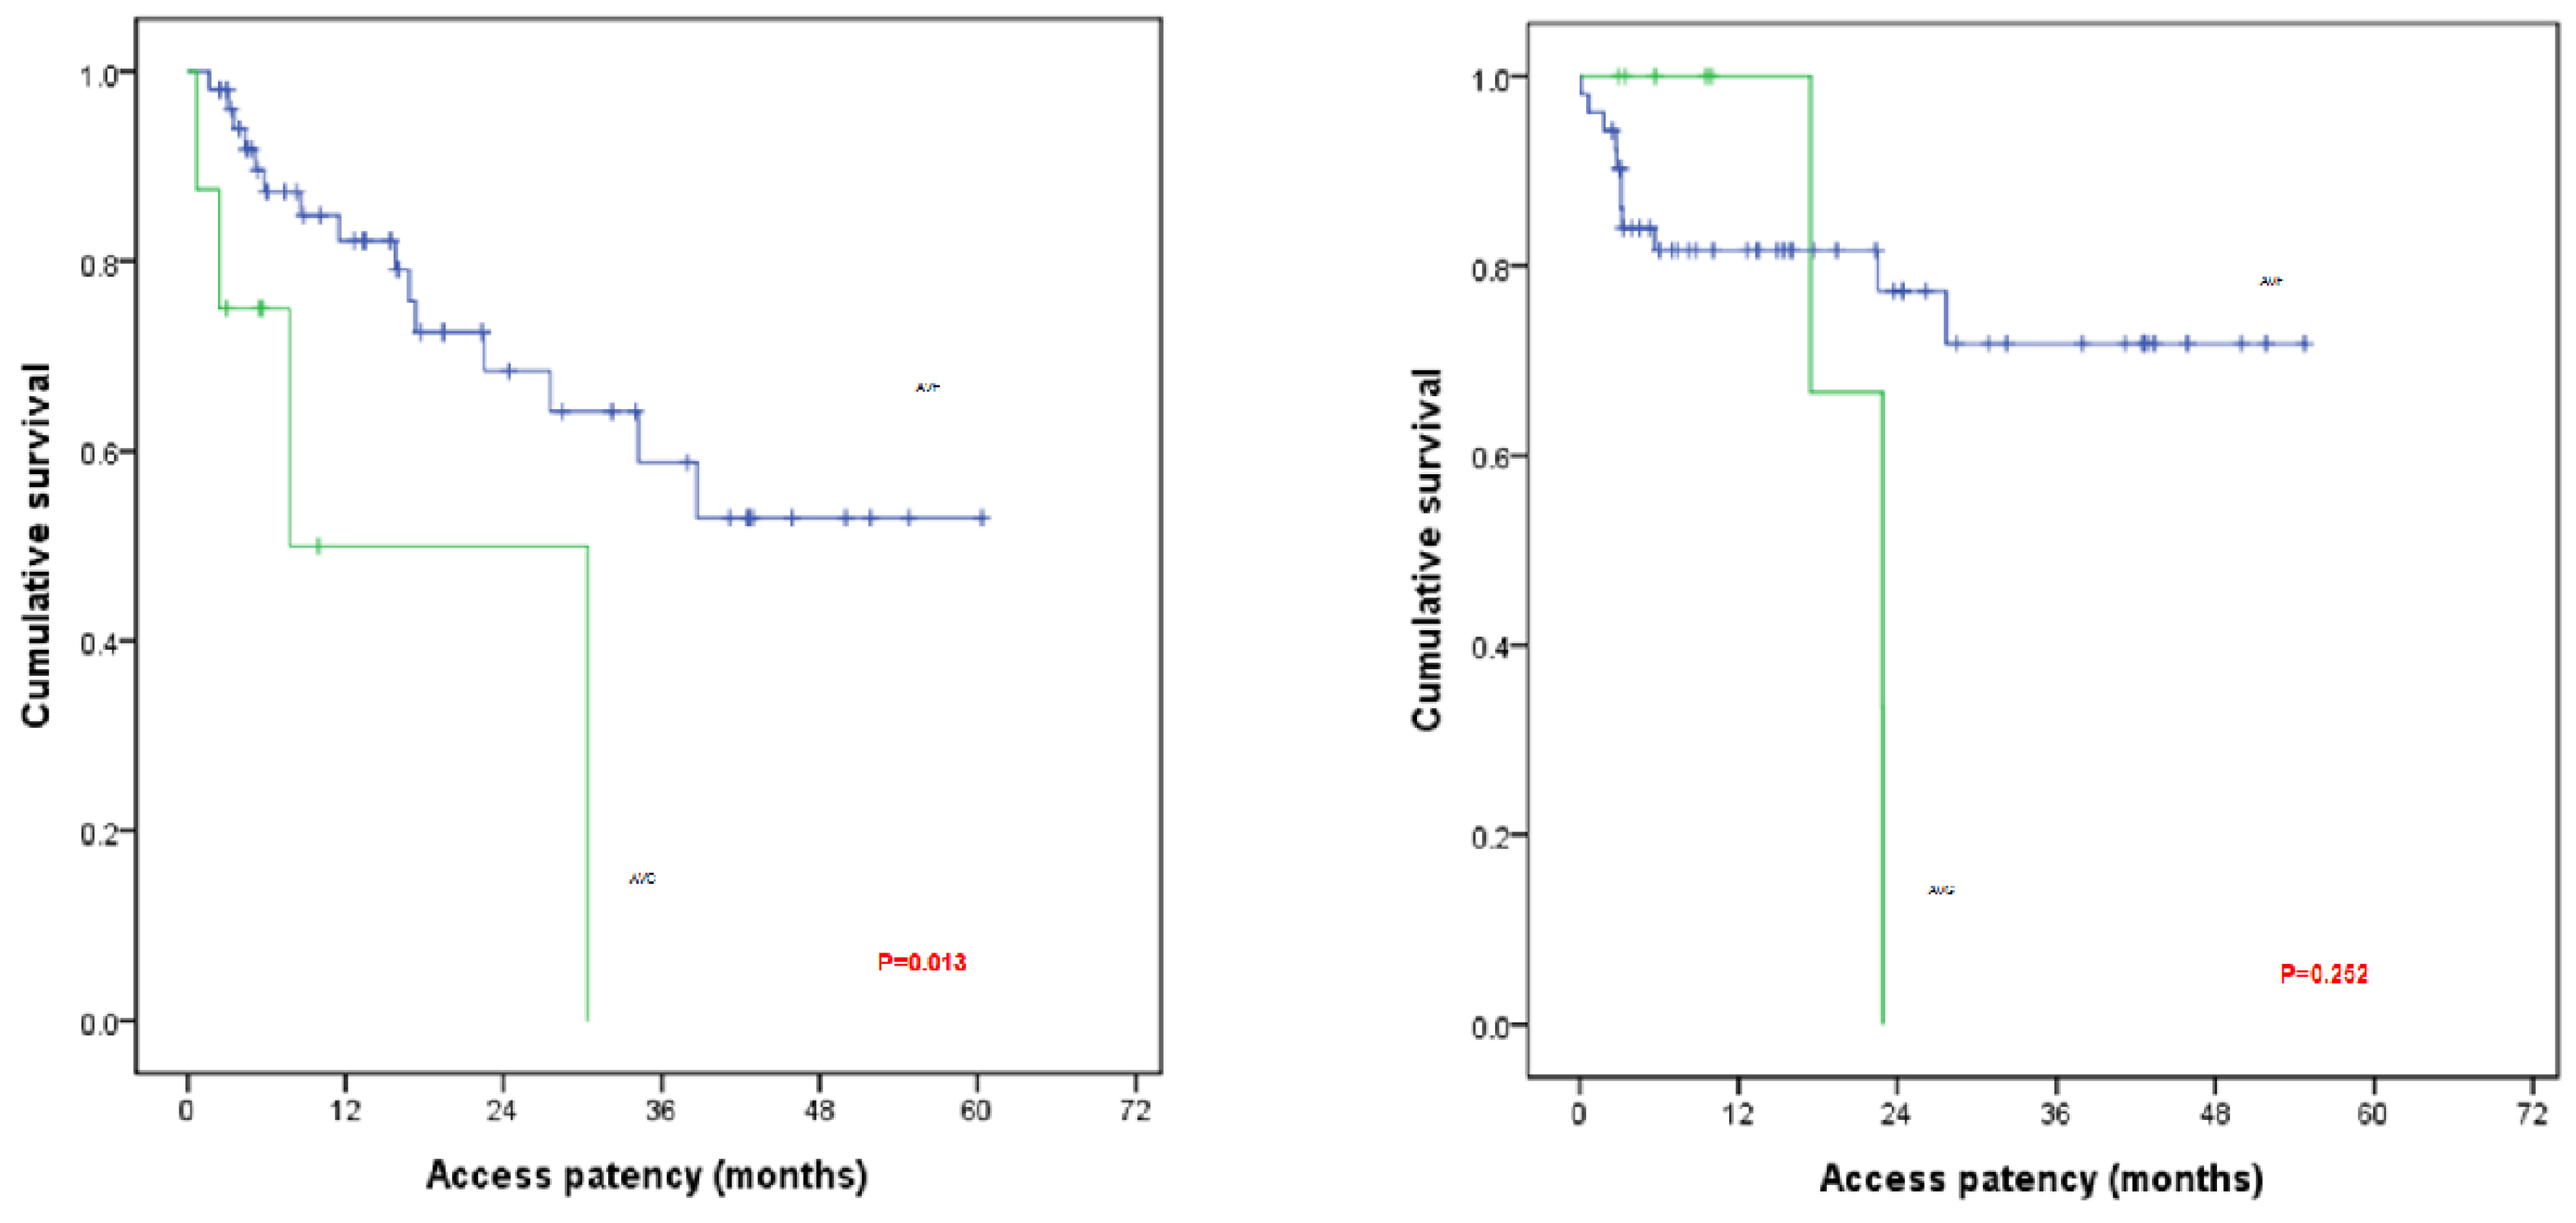
Cumulative survival
Access patency (months)
P=0.013
P=0.252

Figure 2. Primary and Secondary Patency rate

CONCLUSION

In the very elderly patients, the patients with AVF had significantly higher survival rate compared to those with AVG.

KEIMYUNG UNIVERSITY DONGSAN MEDICAL CENTER

53 ERA
533--SP
Dialysis. Vascular access.
Woo Yeong
DOI: 10.3252/pso.eu.53era.2016

ePosters supported by F. Hoffmann- La Roche Ltd.
53rd CONGRESS ERA-EDTA VIENNA AUSTRIA MAY 21st-24th 2016
PosterSessionOnline.com