BSH2020-PO-161

BSH 2020 VIRTUAL

9 -14 NOVEMBER

60 1960–2020 British Society for Haematology Listening • Learning • Leading

Bone and joint health markers in persons with haemophilia A (PwHA) treated with emicizumab▼ in HAVEN 3

A. KIIALAINEN,[1] M. NIGGLI,[1] C. KEMPTON,[2] G. CASTAMAN,[3] T. CHANG,[4] I. PAZ-PRIEL,[4] J. I. ADAMKEWICZ,[4] G. LEVY*[4]

1 F. Hoffmann-La Roche Ltd, Basel, Switzerland; 2 Emory University School of Medicine, Atlanta, GA, USA; 3 Careggi University Hospital, Florence, Italy; 4 Genentech, Inc., South San Francisco, CA, USA;
*GL new affiliation: Spark Therapeutics Inc., Philadelphia, PA, USA

SUMMARY

- Haemophilic arthropathy is a common complication of haemophilia A (HA).[1]
- Treatment of HA with emicizumab was shown to be efficacious and well tolerated in the HAVEN clinical trials.[2–5]
- The effect of emicizumab prophylaxis on bone and joint health was assessed as an exploratory endpoint in HAVEN 3.
- Treatment with emicizumab led to clinically relevant improvements in joint function in those with target joints. Bone and joint biomarkers remained within the normal range during emicizumab prophylaxis.

BACKGROUND

- PwHA are predisposed to recurrent joint bleeds and haemophilic arthropathy which can cause permanent joint damage.[1]
- HA is associated with decreased bone mineral density.[6]
- Emicizumab is a recombinant, bispecific, humanised monoclonal antibody that bridges factor (F) IXa and FX to replace the function of missing FVIIIa in PwHA, restoring haemostasis.[7]
- In HAVEN 3 (NCT02847637), the risk of treated bleeds was significantly reduced compared with episodic FVIII with 96% and 97% reductions (p<0.001) with prophylaxis once weekly and once every 2 weeks (QW, Q2W)), respectively; treatment with emicizumab was well tolerated.[2]
- Here we explore the effect of emicizumab prophylaxis on bone and joint health in PwHA without FVIII inhibitors enrolled in the HAVEN 3 trial.

METHODS

HAVEN 3: A phase III trial of adult and adolescent persons with severe HA without FVIII inhibitors, who previously received FVIII as prophylaxis or episodic treatment (Figure 1).

Figure 1. HAVEN 3 study design.[2]

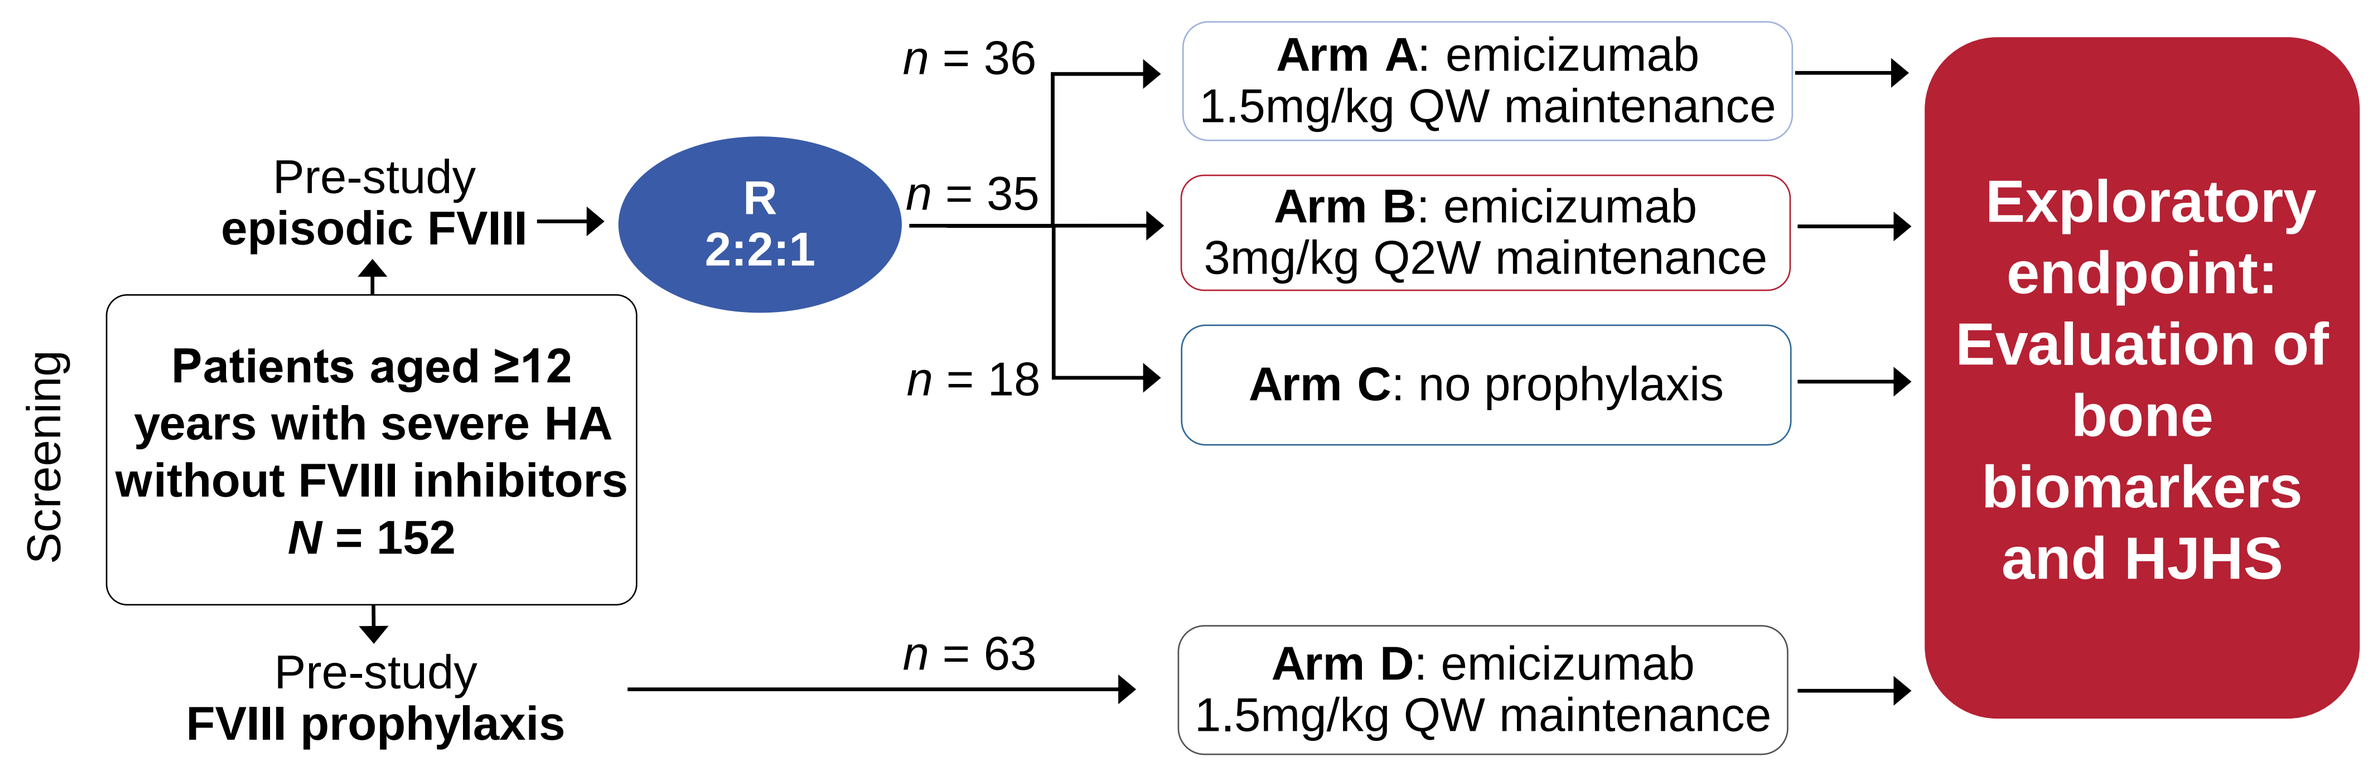
Screening
Patients aged ≥12 years with severe HA without FVIII inhibitors N = 152
Pre-study episodic FVIII
R 2:2:1
n = 36
Arm A: emicizumab 1.5mg/kg QW maintenance
n = 35
Arm B: emicizumab 3mg/kg Q2W maintenance
n = 18
Arm C: no prophylaxis
Pre-study FVIII prophylaxis
n = 63
Arm D: emicizumab 1.5mg/kg QW maintenance
Exploratory endpoint: Evaluation of bone biomarkers and HJHS

All patients treated with emicizumab were given a loading dose of 3mg/kg weekly for 4 weeks, followed by the indicated maintenance dose, starting Week 5. The primary efficacy analysis was performed at 24 weeks; emicizumab prophylaxis continued after the primary efficacy analysis.
FVIII, factor VIII; HA, haemophilia A; HJHS, Haemophilia Joint Health Score; QW, every week; Q2W, every 2 weeks; R, randomisation.

- Haemophilia joint health scores (HJHS; v2.1) were evaluated at baseline and Week 49 of emicizumab prophylaxis in 107 PwHA.
 - Higher HJHS scores indicate poorer joint health.
- Biomarkers of bone formation (osteocalcin, N-terminal propeptide of type I procollagen), bone resorption (C-terminal telopeptide of type I collagen), osteoblasts (osteoprotegerin), osteoclastogenesis (soluble receptor activator of nuclear factor- kappaB Ligand), cartilage turnover (cartilage oligomeric matrix protein), cartilage degradation (C-terminal telopeptide of type II collagen), cartilage synthesis/repair (a chondroitin sulfate epitope – CS-846), and inflammation (interleukin [IL] 1 beta, IL-6, and tumour necrosis factor alpha) were measured in 117 PwHA at baseline and after 13, 25, 49, and 73 weeks of emicizumab prophylaxis.
- In total, 78 PwHA were part of both the biomarker analysis and HJHS evaluation.

RESULTS

Biomarker and HJHS data were analysed from PwHA in HAVEN 3 (Table 1).

- At data cut-off (4 October 2018), median follow-up was 87.4 weeks.

Table 1. Participant characteristics.

Characteristics		PwHA with evaluable HJHS (*n* = 107)	PwHA with biomarker data (*n* = 117)
Median age (IQR), years		35 (26–45)	38 (27–48)
Age groups, *n* (%)	≥65 years	2 (1.9)	5 (4.3)
	<18 years	7 (6.5)	7 (6.0)
Median BMI (IQR), kg/m²		25.0 (23.0–28.0)	25.4 (22.8–28.3)
Race, *n* (%)	White	64 (59.8)	80 (68.4)
	Asian	28 (26.2)	22 (18.8)
	Black/African American	5 (4.7)	5 (4.3)
	Other	1 (0.9)	1 (0.8)
	Unknown	9 (8.4)	9 (7.7)
Prior FVIII, *n* (%)	Prophylaxis	47 (43.9)	50 (42.7)
	Episodic	60 (56.1)	67 (57.3)
Target joints, *n* (%)	None	36 (33.6)	38 (32.5)
	≥1	71 (66.4)	79 (67.5)
History of HIV infection, *n* (%)		17 (15.0)	31 (26.5)
History of osteoporosis, *n* (%)	Any	1 (0.9)	5 (4.3)
	Treated	1 (0.9)	4 (3.4)

BMI, body mass index; FVIII, factor VIII; HIV, human immunodeficiency virus;
HJHS, Haemophilia Joint Health Score; IQR, interquartile range;
PwHA, persons with haemophilia A; QW, every week.

PwHA previously on FVIII prophylaxis and with no target joints had lower (indicating healthier) HJHS scores at baseline (Figure 2).

Figure 2. HJHS scores ≥/<12 at baseline by treatment (A) or target joint status (B).

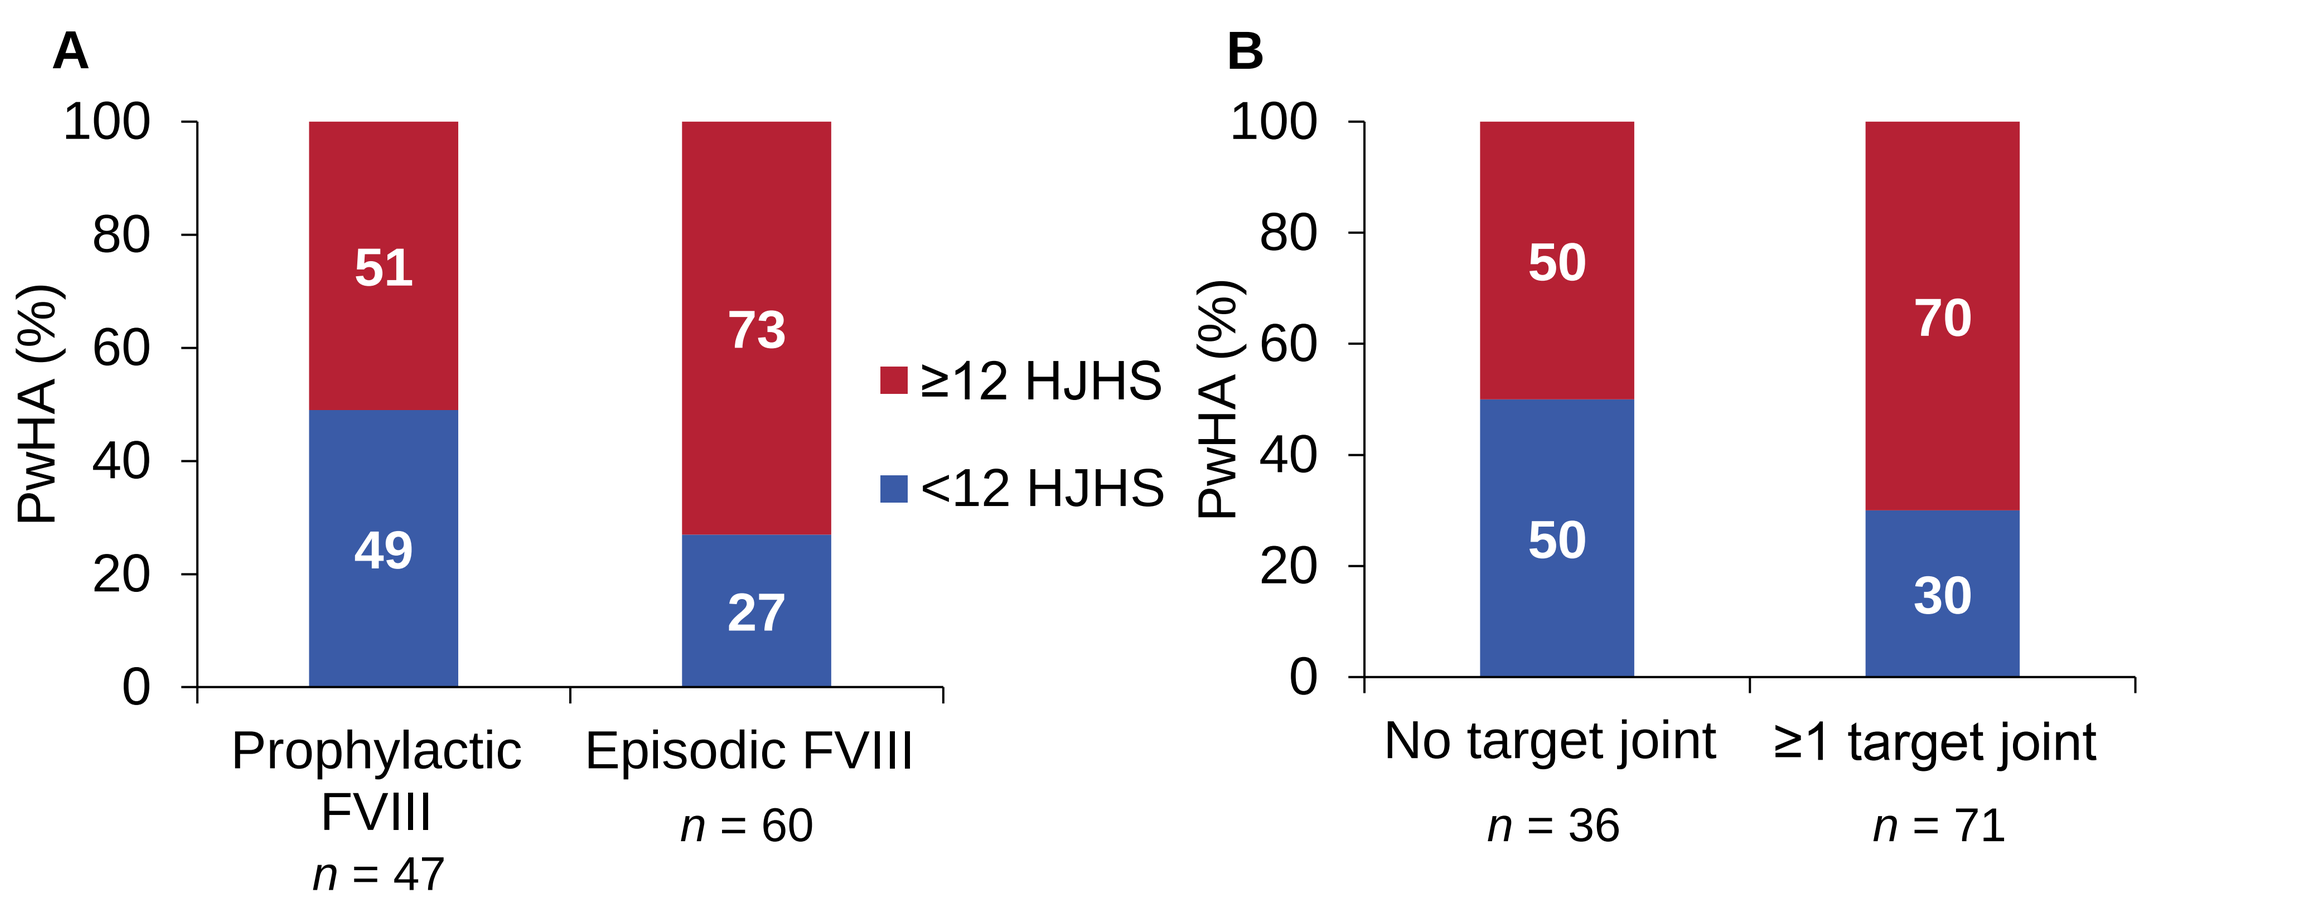
A
PwHA (%)
100
80
60
40
20
0
51
49
73
27
≥12 HJHS
<12 HJHS
Prophylactic FVIII n = 47
Episodic FVIII n = 60
B
50
50
70
30
No target joint n = 36
≥1 target joint n = 71

The HJHS 2.1 consists of 8 item scores on joint level and a global gait score. Scores range from 0 to 20 per joint and the global gait score ranges from 0 to 4, resulting in a HJHS-total score (0 to 124).
A cut-off score of 12 was used (median in the FVIII prophylaxis population based on our data).
FVIII, factor VIII; HJHS, Haemophilia Joint Health Score;
PwHA, persons with haemophilia A.

Significant and clinically relevant improvements in the HJHS joint-specific domain were observed at 48 weeks in PwHA with ≥1 target joint.

- Mean change in joint-specific and total HJHS after 48 weeks of emicizumab prophylaxis was -2.23 and -2.25, respectively, for PwHA with ≥1 target joint (**Figure 3**).
- Improvements were consistent across HJHS for different locations (knee, ankle, elbow).

Figure 3. Improvement in joint health at Week 49 of emicizumab prophylaxis.

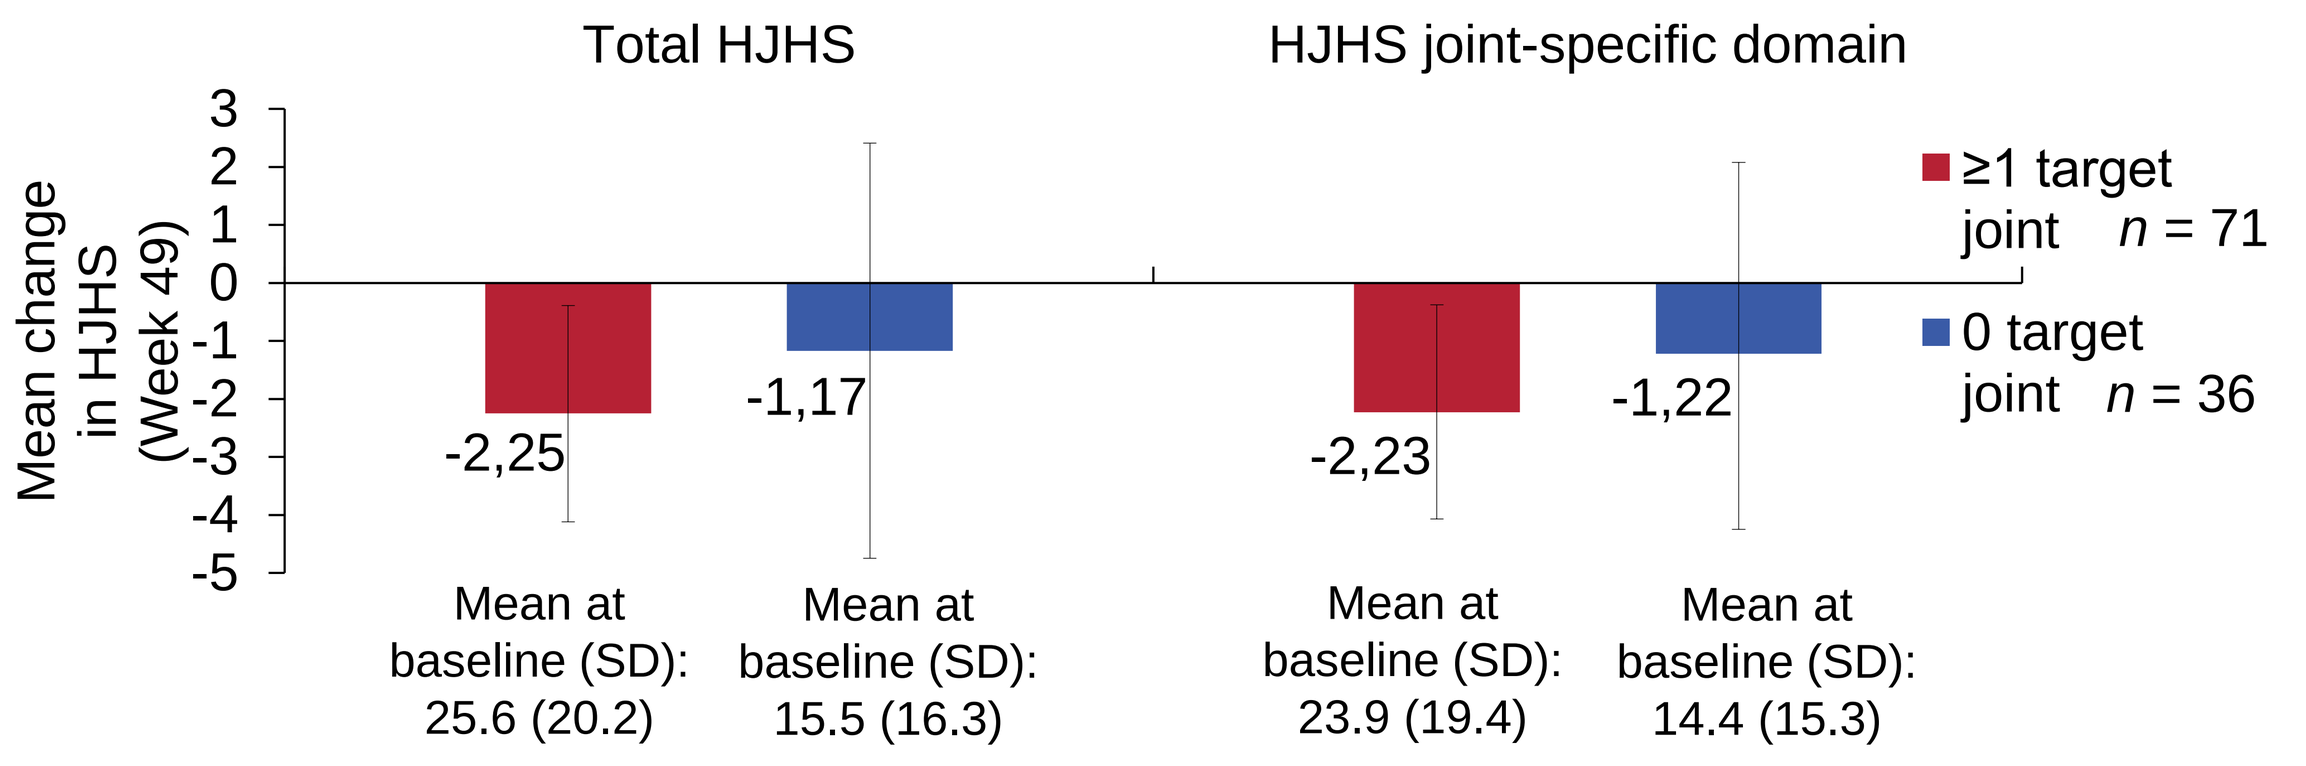
Total HJHS
HJHS joint-specific domain
Mean change in HJHS (Week 49)
3
2
1
0
-1
-2
-3
-4
-5
-2,25
-1,17
-2,23
-1,22
≥1 target joint n = 71
0 target joint n = 36
Mean at baseline (SD): 25.6 (20.2)
Mean at baseline (SD): 15.5 (16.3)
Mean at baseline (SD): 23.9 (19.4)
Mean at baseline (SD): 14.4 (15.3)

Clinically relevant improvements in HJHS are defined as a ≥2-point reduction in the joint-specific domain or a ≥4-point reduction in total HJHS.[8]
Results were significant in the exploratory sense with 95% confidence interval (shown as error bars) not including 0. HJHS was measured at the start of the study and at Week 49.
Data exclude Arm C and include only those with evaluable HJHS score at both baseline and Week 49.
HJHS, Haemophilia Joint Health Score; SD, standard deviation.

Baseline values of most bone and joint biomarkers were within normal ranges or similar to published levels in healthy individuals.[9–12]

- Large variability in bone and joint biomarkers was observed between individuals (**Figure 4**).
- No significant differences in baseline values were observed in the biomarkers of PwHA previously on FVIII prophylaxis versus episodic treatment, and in PwHA with target joints versus those without.

Figure 4. Baseline biomarker levels.

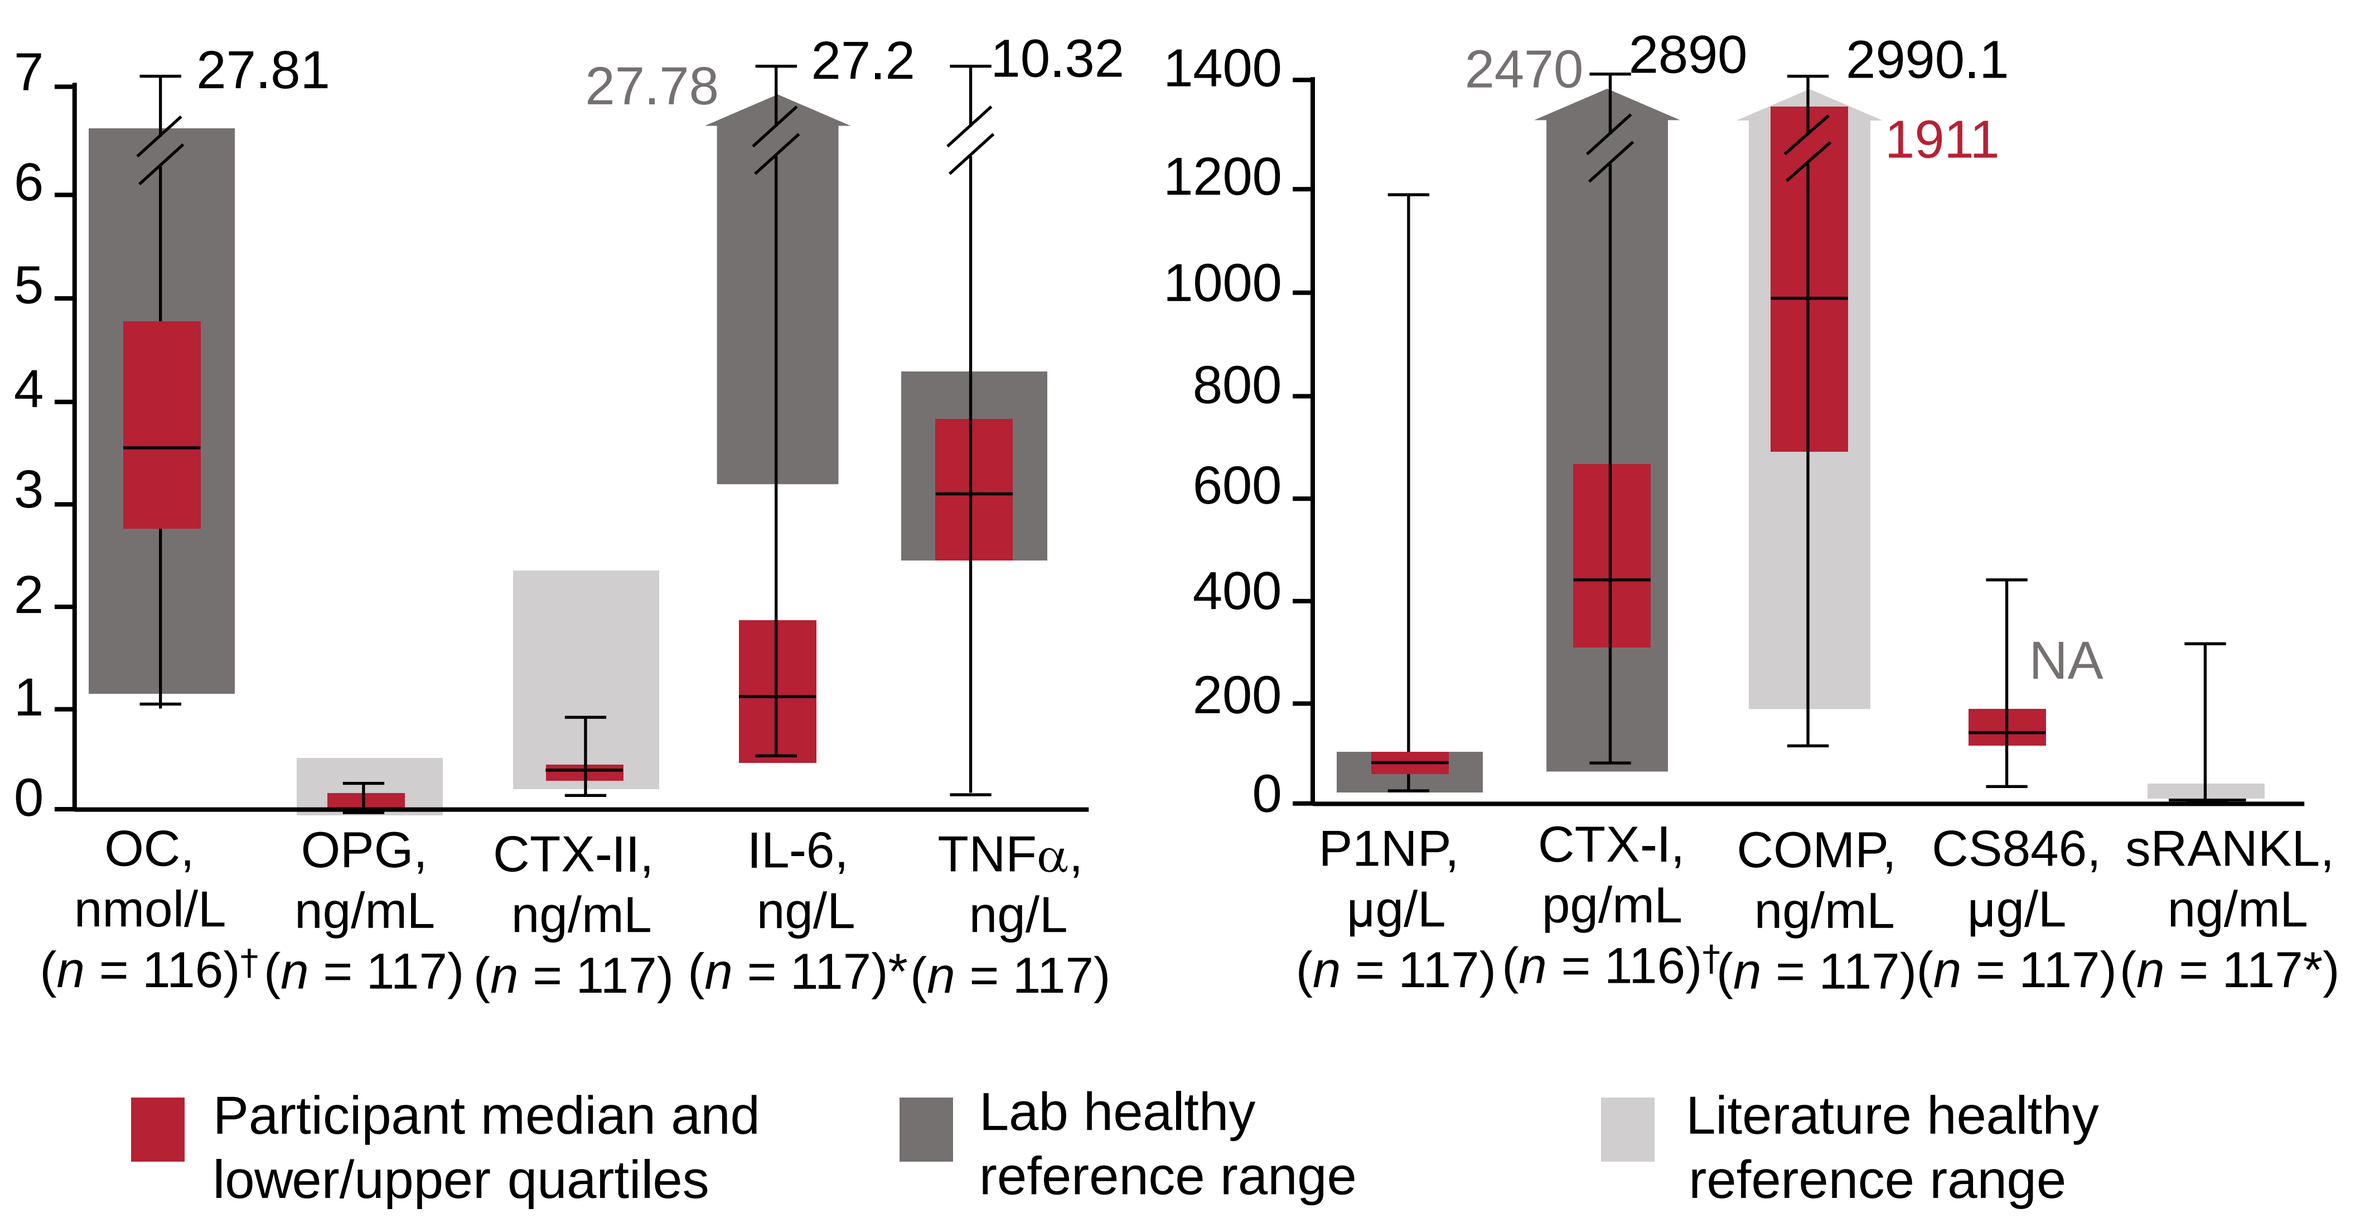
27.81
27.78
27.2
10.32
2470
2890
2990.1
1911
NA
OC, nmol/L (n = 116)†
OPG, ng/mL (n = 117)
CTX-II, ng/mL (n = 117)
IL-6, ng/L (n = 117)*
TNFα, ng/L (n = 117)
P1NP, µg/L (n = 117)
CTX-I, pg/mL (n = 116)†
COMP, ng/mL (n = 117)
CS846, µg/L (n = 117)
sRANKL, ng/mL (n = 117*)
Participant median and lower/upper quartiles
Lab healthy reference range
Literature healthy reference range

*Values below the limit of quantification (BLQ) are imputed with half of that limit of quantification; 57% of values for sRANKL, and 49% of values for IL-6 were BLQ; all IL-1β samples but one were BLQ and so are not shown here.
†Missing observation at baseline for *n* = 1.
Error bars denote min/max values.
COMP, cartilage oligomeric matrix protein; CS-846, chondroitin sulfate 846 epitope;
CTXI, C-terminal telopeptide of type I collagen; CTX-II, C-terminal telopeptide of type II collagen;
IL-6, interleukin 6; OC, osteocalcin; OPG, osteoprotegerin; P1NP, N-terminal propeptide of type I procollagen; sRANKL, soluble receptor activator of nuclear factor- kappaB Ligand;
TNFα, tumour necrosis factor alpha.

None of the measured biomarkers changed significantly during emicizumab prophylaxis across 18 months (Figure 5).

- During the follow-up period, all biomarker levels (e.g., COMP and P1NP; *n* = 94) remained stable within the normal range.

Figure 5. Measures of cartilage turnover (A. COMP) and bone formation (B. P1NP).

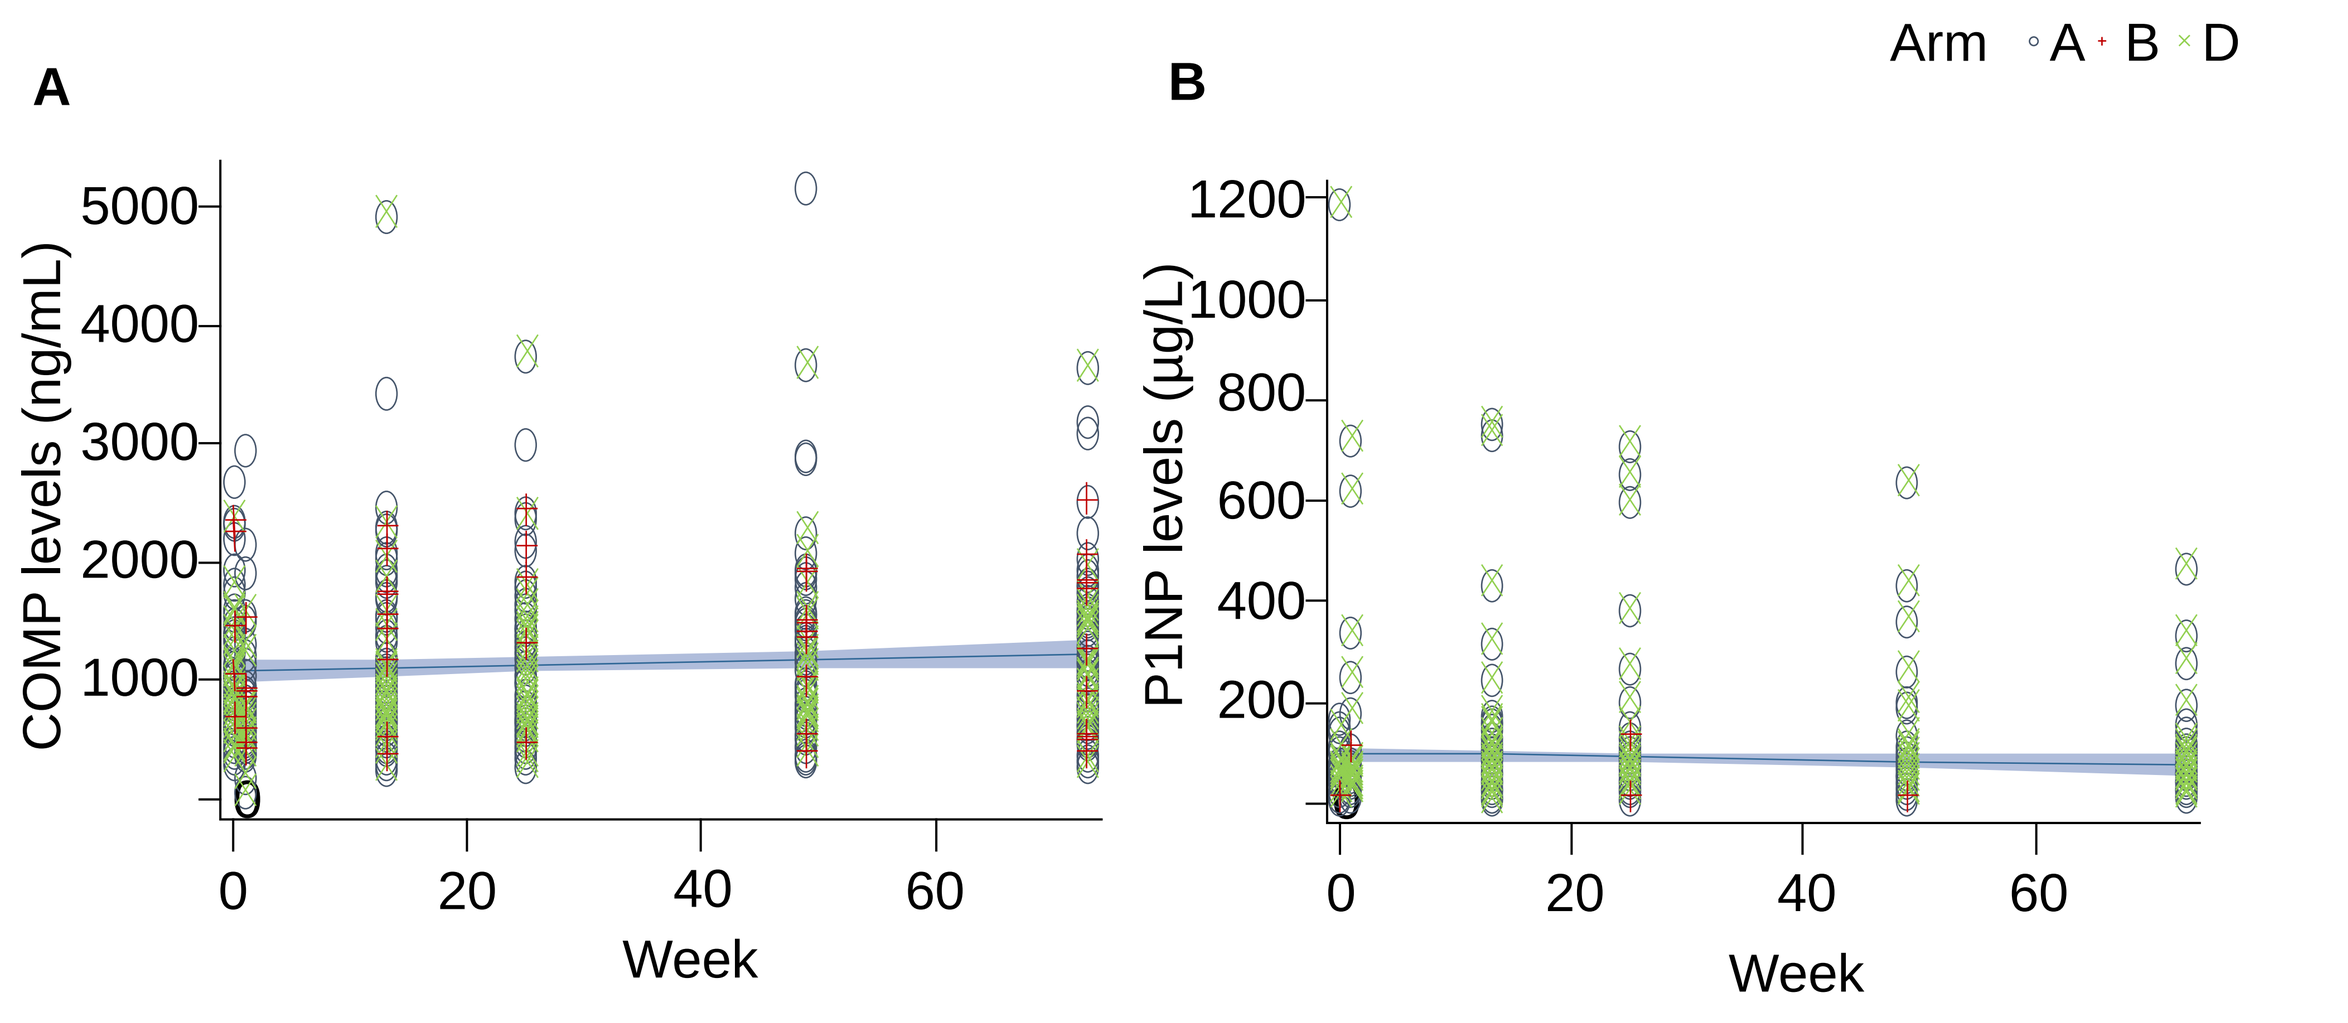
Arm A B D
A
COMP levels (ng/mL)
B
P1NP levels (µg/L)
Week
0
20
40
60

Includes only participants ≥18 years, and excludes Arm C.
COMP, cartilage oligomeric matrix protein; P1NP, N-terminal propeptide of type I procollagen.

CONCLUSIONS

- Clinically relevant improvements in HJHS were observed in PwHA with target joints after as little as 48 weeks of emicizumab.
- At baseline, bone and joint biomarker levels were similar to those reported in healthy individuals, and remained within normal ranges during the follow-up period; as levels were already similar to those reported in healthy individuals, there was little possibility for improvement.
- There was no evidence of worsening in any bone and joint health markers in PwHA on emicizumab where FVIII exposure was reduced.

ACKNOWLEDGEMENTS

The authors would like to thank the study participants and their families, as well as the study investigators and site personnel. This study was co-sponsored by F. Hoffmann-La Roche Ltd and Chugai Pharmaceutical Co., Ltd. Editorial assistance was provided by Sam Brown, MSc and Alex Coulthard, BSc, of Gardiner-Caldwell Communications and was funded by Chugai Pharma UK Ltd.

REFERENCES

1. **Melchiorre, D, et al.** *J Clin Med* 2017;6:63; 2. **Mahlangu J, et al.** *N Engl J Med* 2018;379:811–22; 3. **Oldenburg J, et al.** *N Engl J Med* 2017;377:809–18;
4. **Young G, et al.** *Blood* 2019;134:2127–38; 5. **Pipe SW, et al.** *Lancet Haematol* 2019;6:e295–e305; 6. **Kempton CL, et al.** *Haemophilia* 2014;20:121–8;
7. **Kitazawa T, et al.** *Thromb Haemost* 2017;117:1348–57; 8. **Kuijlaars IAR, et al.** *Haemophilia* 2017;23:934–40; 9. **Kudlacek S, et al.** *Bone* 2003;32:681–86;
10. **Papassava M, et al.** *Epilepsy Behav.* 2019;97:192–6; 11. BioVendor. COMP ELISA insert; 12. **Severino RM, et al.** *Rev Bras Ortop*. 2015;50:331–35.

DISCLOSURES

AK: Employee of and holder of stocks in F. Hoffmann-La Roche Ltd; **MN:** Employee of F. Hoffmann-La Roche Ltd; **CK:** Receipt of research funding from Novo Nordisk; receipt of honoraria from Genentech, Inc., Pfizer and Octapharma; **GC:** Receipt of research funding from Pfizer, CSL Behring and Sobi, consultant for F. Hoffmann-La Roche Ltd, member of speakers' bureau for Bayer, Takeda, CSL Behring, Sobi, Novo Nordisk, Kedrion, Uniqure, F. Hoffmann-La Roche Ltd, Werfen Ltd, Sanofi and Alexion Pharmaceuticals; **TC:** Employee of and holder of stocks in Genentech, Inc.; **IP-P:** Employee of Genentech, Inc.; **JIA:** Employee of and holder of patents/intellectual property in Genentech, Inc., including receipt of royalties; holder of stocks in F. Hoffmann-La Roche Ltd; **GL:** Current employee of Spark Therapeutics Inc., and previous employee of Genentech, Inc.

The data presented here were previously presented at the American Society of Hematology Annual Meeting 2019 (December 7–10, Orlando, Florida).

▼ This medicinal product is subject to additional monitoring. This will allow quick identification of new safety information. Healthcare professionals are asked to report any suspected adverse reactions. These should be reported to the Regulatory Authorities according to UK requirements.

BSH2020 PO-161-B
Thrombosis and Haemostasis
Anna Kiialainen
Poster presented at: British Society for Haematology Listening • Learning • Leading
PosterSessionOnline.com