BSH 2020 VIRTUAL

9 -14 NOVEMBER

60 1960–2020 British Society for Haematology
Listening • Learning • Leading

One-Year Efficacy and Safety From a Phase 3 Trial of Ravulizumab in Adult Patients With Paroxysmal Nocturnal Haemoglobinuria Receiving Prior Eculizumab Treatment

Austin Kulasekararaj[1], Lindsay Mitchell[2] (presenter), Anita Hill[3], Saskia Langemeijer[4], Richard Wells[5], F. Ataúlfo González Fernández[6], Anna Gaya[7], Emilio Ojeda Gutiérrez[8], Caroline Piatek[9], Kensuke Usuki[10], Alberto Bosi[11], Robert Brodsky[12], Masayo Ogawa[13], Ji Yu[13], Stephan Ortiz[13], Alexander Röth[14], Jong Wook Lee[15], Regis Peffault de la Tour[16,17]

[1]Department of Haematological Medicine, King's College Hospital, NIHR/Wellcome King's Clinical Research Facility, London, [2]University Hospital Monklands, North Lanarkshire, [3]Department of Haematology, Leeds Teaching Hospitals, Leeds, United Kingdom, [4]Radboudumc, Nijmegen, Netherlands, [5]Sunnybrook Health Sciences Centre, Toronto, Canada, [6]Department of Hematology, Hospital Clinico Universitario San Carlos, Madrid, [7]Hospital Clinic de Barcelona, Barcelona, [8]Hospital Universitario Puerta de Hierro Majadahonda, Madrid, Spain, [9]Jane Anne Nohl Division of Hematology, Keck School of Medicine, University of Southern California, Los Angeles, CA, United States, [10]NTT Medical Center Tokyo, Tokyo, Japan, [11]Department of Experimental and Clinical Medicine, University of Florence, Florence, Italy, [12]The Johns Hopkins University School of Medicine, Baltimore, MD, [13]Alexion Pharmaceuticals, Inc., Boston, MA, United States, [14]Department of Hematology, West German Cancer Center, University Hospital Essen, Essen, Germany, [15]Department of Hematology, Seoul St. Mary's Hospital, The Catholic University of Korea, Seoul, Korea, Republic Of, [16]Bone Marrow Transplantation Unit, Saint Louis Hospital, Assistance Publique–Hôpitaux de Paris, [17]French Reference Center for Aplastic Anemia and Paroxysmal Nocturnal Hemoglobinuria, Saint Louis Hospital and University Paris Diderot, Paris, France

INTRODUCTION

- Ravulizumab is an innovative long-acting C5 complement inhibitor approved in the United States,[1] Europe,[2] Japan,[3] Canada,[4] Brazi,[5] and Australia[6] for the treatment of paroxysmal nocturnal haemoglobinuria (PNH)
- In the largest phase 3 study (ALXN1210-PNH-302; NCT03056040) in eculizumab-experienced patients with PNH, ravulizumab administered once every 8 weeks (q8w) was shown to be noninferior to eculizumab administered once every 2 weeks (q2w) after 26 weeks for all primary and key secondary endpoints7
- At the end of the randomised evaluation period (26 weeks), all patients had the option to receive weight-based dosing of ravulizumab in an extension of ALXN1210-PNH-302

OBJECTIVES

- Characterise the durability of responses to ravulizumab through 52 weeks of treatment
- Evaluate the efficacy profile of ravulizumab in adult patients with PNH after switching from stable eculizumab therapy
- Evaluate serum free C5 levels
- Evaluate the safety profile in each treatment arm

METHODS

Study Design and Patients

- This was an extension of the open-label, phase 3, multicenter study ALXN1210-PNH-302 (NCT03056040; **Figure 1**)
- Adult patients with PNH, stable on eculizumab for ≥ 6 months, and with lactate dehydrogenase (LDH) ≤ 1.5 times the upper limit of normal (ULN) at screening were randomly assigned 1:1 to switch to ravulizumab or to continue receiving eculizumab
- Patients randomised to ravulizumab received weight-based loading (day 1)/maintenance doses (day 15 and q8w thereafter)
 - ≥ 40 to < 60 kg body weight: 2400/3000 mg
 - ≥ 60 to < 100 kg: 2700/3300 mg
 - ≥ 100 kg: 3000/3600 mg
- Patients randomised to eculizumab continued receiving 900 mg q2w
- After 26 weeks, patients in the ravulizumab arm continued ravulizumab maintenance therapy (R-R arm), while patients in the eculizumab arm were then switched to ravulizumab (E-R arm)

Figure 1. Study Design

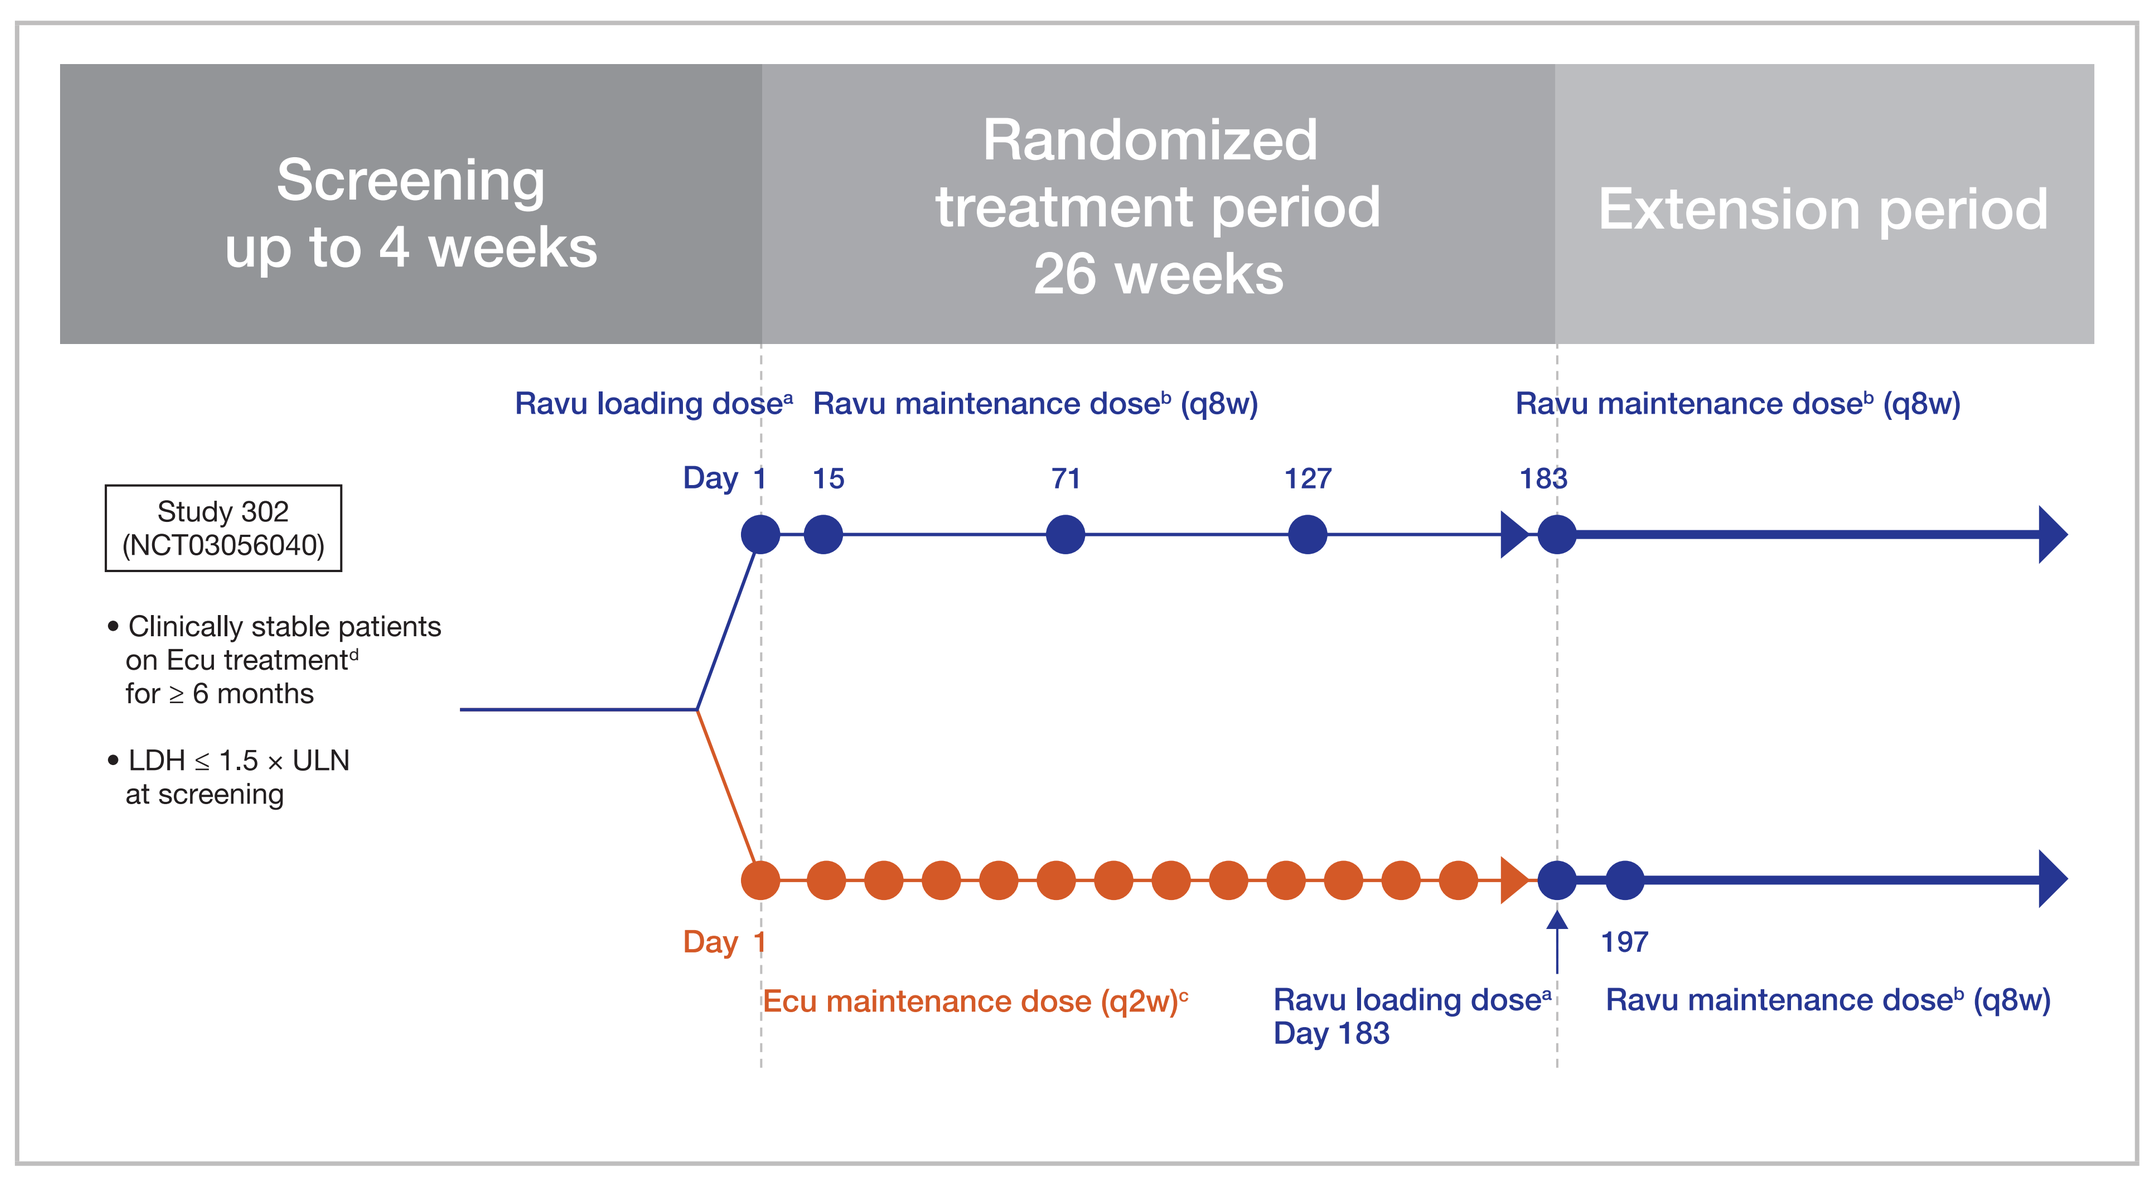

Screening up to 4 weeks
Randomized treatment period 26 weeks
Extension period
Ravu loading dose[a] Ravu maintenance dose[b] (q8w)
Ravu maintenance dose[b] (q8w)
Day 1 15 71 127 183
Study 302 (NCT03056040)
• Clinically stable patients on Ecu treatment[d] for ≥ 6 months
• LDH ≤ 1.5 × ULN at screening
Day 1
197
Ecu maintenance dose (q2w)[c]
Ravu loading dose[a] Day 183
Ravu maintenance dose[b] (q8w)

Ecu, eculizumab; LDH, lactate dehydrogenase; q2w, once every 2 weeks; q8w, once every 8 weeks; Ravu, ravulizumab; ULN, upper limit of normal.

[a] Ravu loading dose: 2400 mg for patients weighing ≥ 40 to < 60 kg, 2700 mg for patients weighing ≥ 60 to < 100 kg, 3000 mg for patients weighing ≥ 100 kg.

[b] Ravu maintenance dose: 3000 mg for patients weighing ≥ 40 to < 60 kg, 3300 mg for patients weighing ≥ 60 to < 100 kg, 3600 mg for patients weighing ≥ 100 kg.

[c] Ecu maintenance dose: 900 mg.

[d] Approved dose for PNH.

Endpoints

- The primary efficacy endpoint was percent change in LDH from baseline
- Key secondary efficacy endpoints included proportion of patients with breakthrough haemolysis (BTH), transfusion avoidance (TA), stabilised hemoglobin (HGB-S) levels, and improvement in FACIT-Fatigue total score
- Other endpoints included safety and change in serum free C5 levels from baseline
 - Serum free C5 < 0.5 µg/mL correlated with maximal intravascular control and complete terminal complement inhibition[1,2]

CONCLUSIONS

- Adult patients with PNH on stable eculizumab therapy who received ravulizumab over 52 weeks (R-R arm) experienced durable efficacy
- Adult patients with PNH on stable eculizumab therapy who received eculizumab for 26 weeks and then switched to ravulizumab (E-R arm) had an efficacy response consistent with patients in the R-R arm
- All patients receiving eculizumab who had suboptimal free C5 inhibition achieved and maintained complete free C5 inhibition after the switch to ravulizumab
- No BTH events were associated with elevated serum free C5 levels
- Ravulizumab continued to be well tolerated through week 52 with no new safety concerns

RESULTS

Patients

- All 191 patients who completed treatment entered the extension period (Figure 2)
- At 52 weeks, 189 patients were being treated with ravulizumab, 1 patient discontinued due to physician's decisions, and 1 patient withdrew from the study

Figure 2. Patient Flow Through Week 52

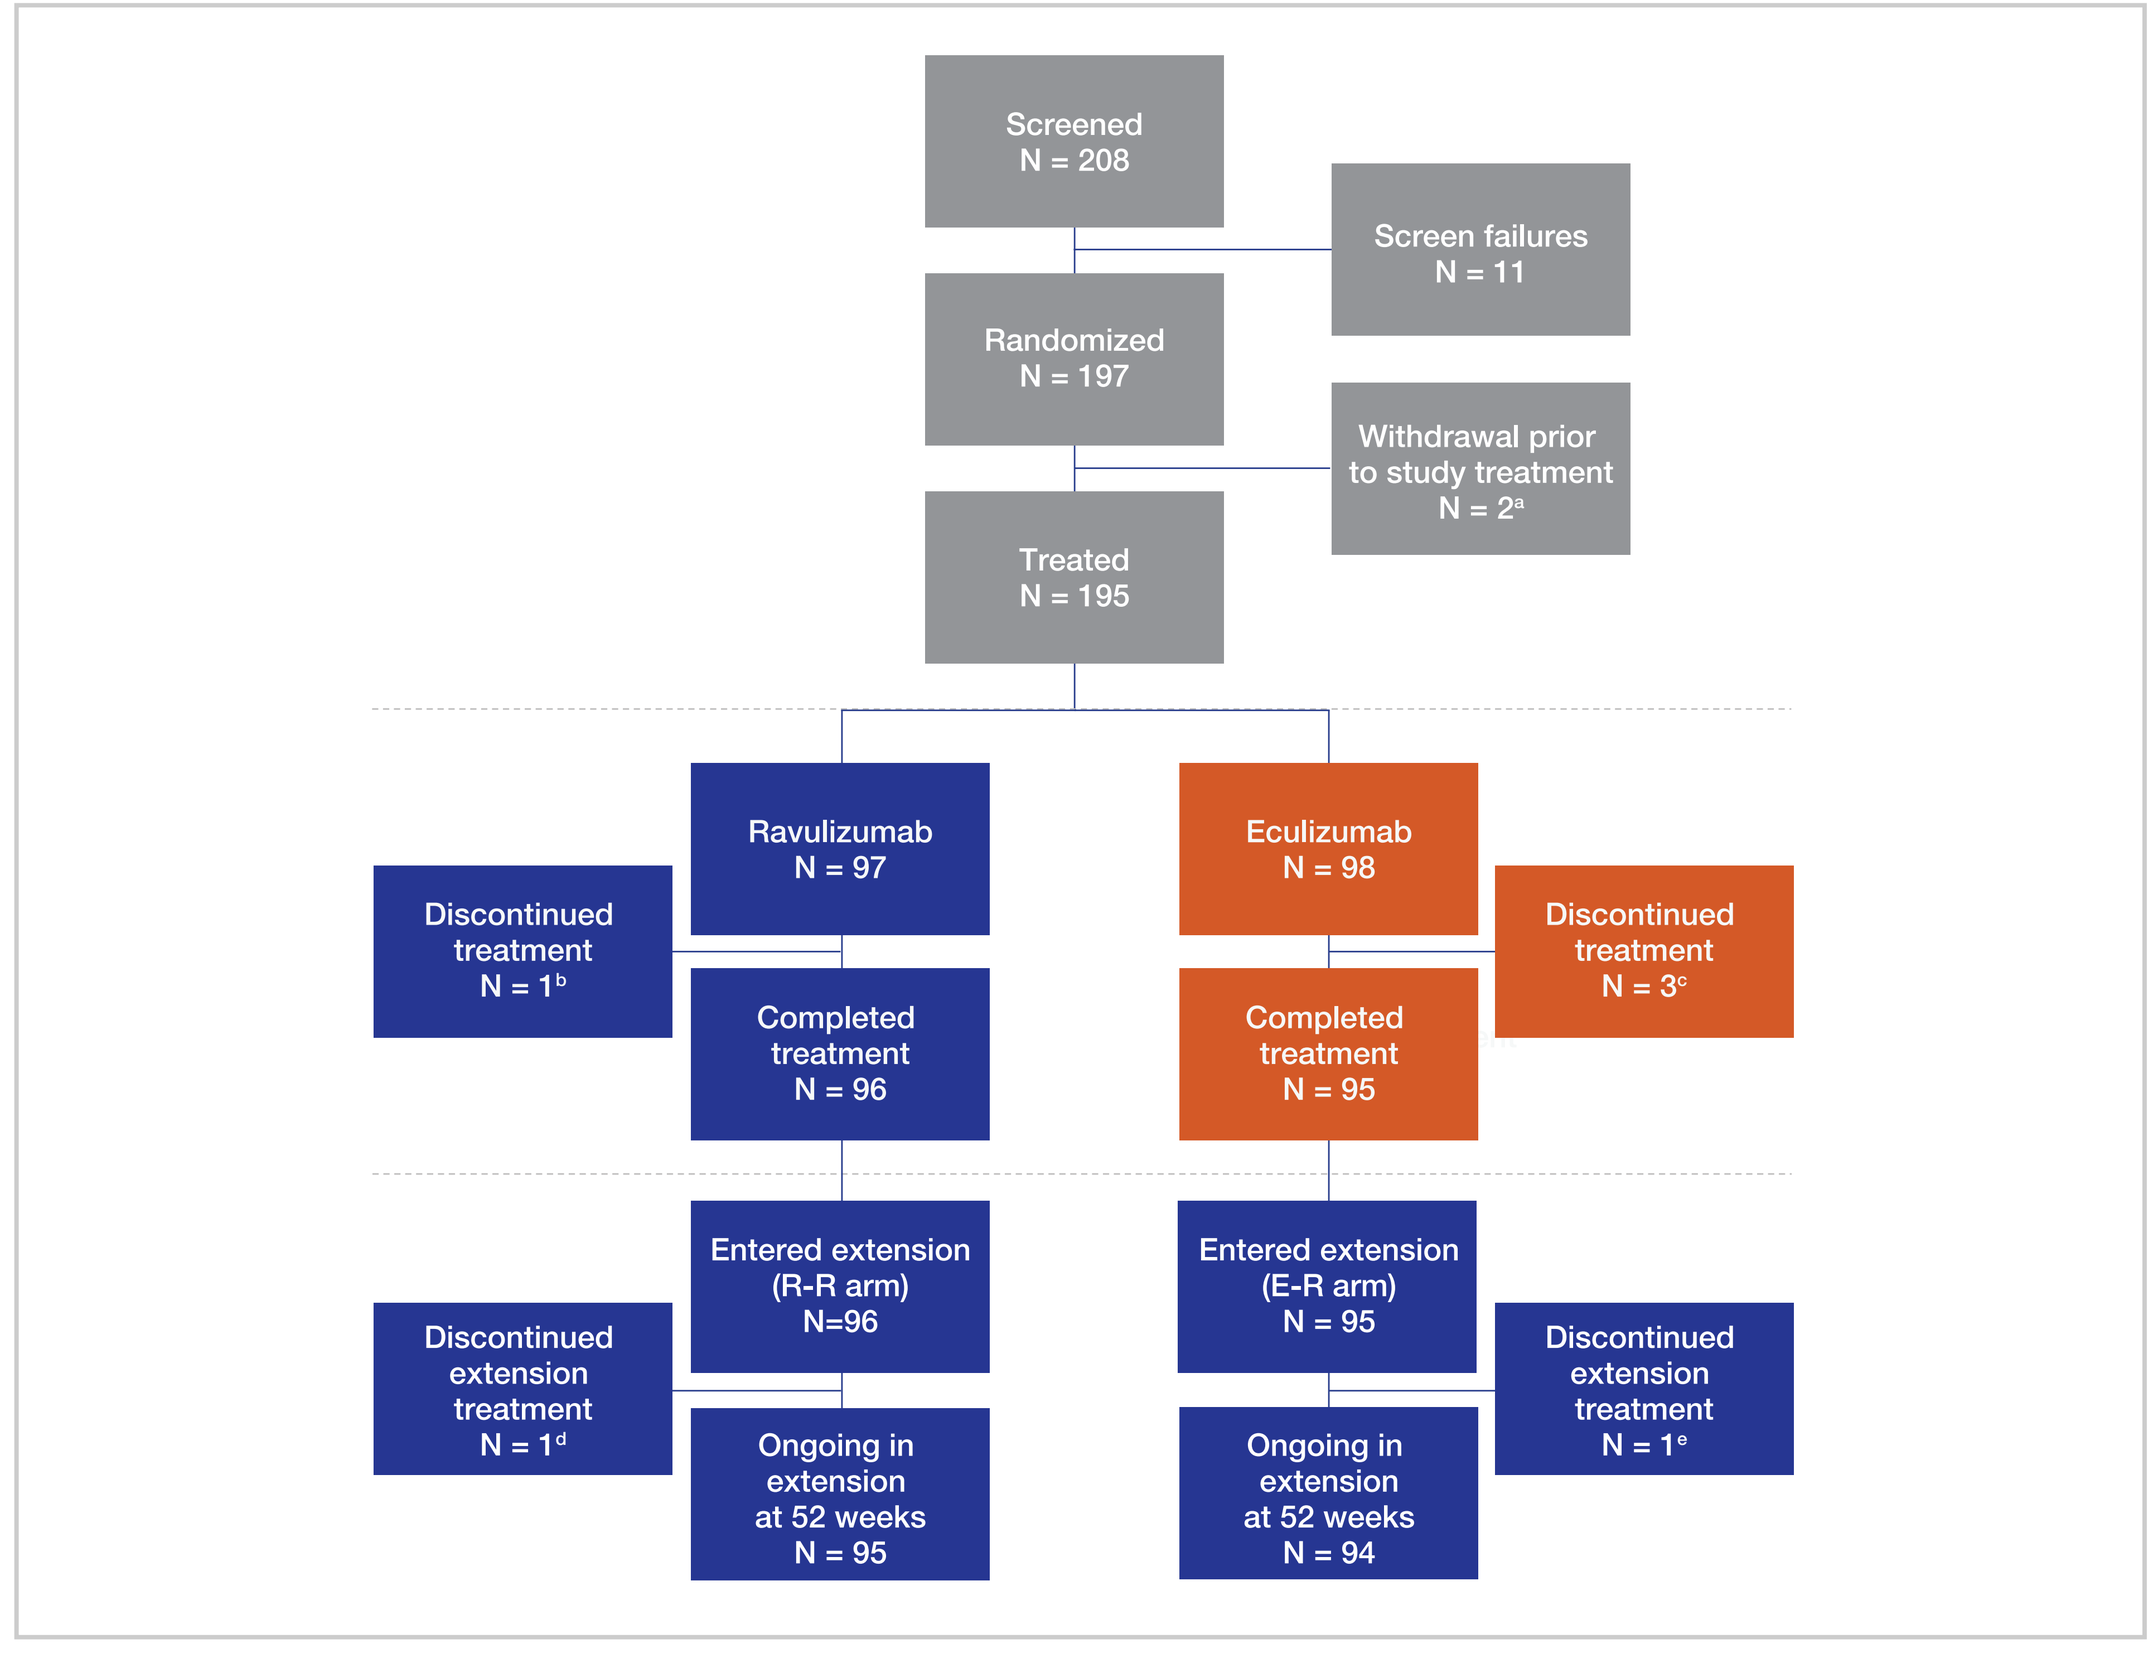

Screened N = 208
Screen failures N = 11
Randomized N = 197
Withdrawal prior to study treatment N = 2[a]
Treated N = 195
Ravulizumab N = 97
Eculizumab N = 98
Discontinued treatment N = 1[b]
Discontinued treatment N = 3[c]
Completed treatment N = 96
Completed treatment N = 95
Entered extension (R-R arm) N=96
Entered extension (E-R arm) N = 95
Discontinued extension treatment N = 1[d]
Discontinued extension treatment N = 1[e]
Ongoing in extension at 52 weeks N = 95
Ongoing in extension at 52 weeks N = 94

Primary Efficacy Endpoint

Percentage Change in LDH from Baseline

- Patients in both groups showed a durable response for percentage change in LDH up to 52 weeks, similar to what was observed during the first 26 weeks (**Figure 3**)
- At 52 weeks, patients in the R-R arm had an 8.8% (SD, 29%) increase in LDH from baseline, while patients in the E-R arm had a 5.8% (27%) increase in LDH from baseline
- Mean LDH levels in both groups were maintained at 1.0 x ULN (< 246 U/L)

Figure 3. Mean (95% CI) Percentage Change in Lactate Dehydrogenase Over Time

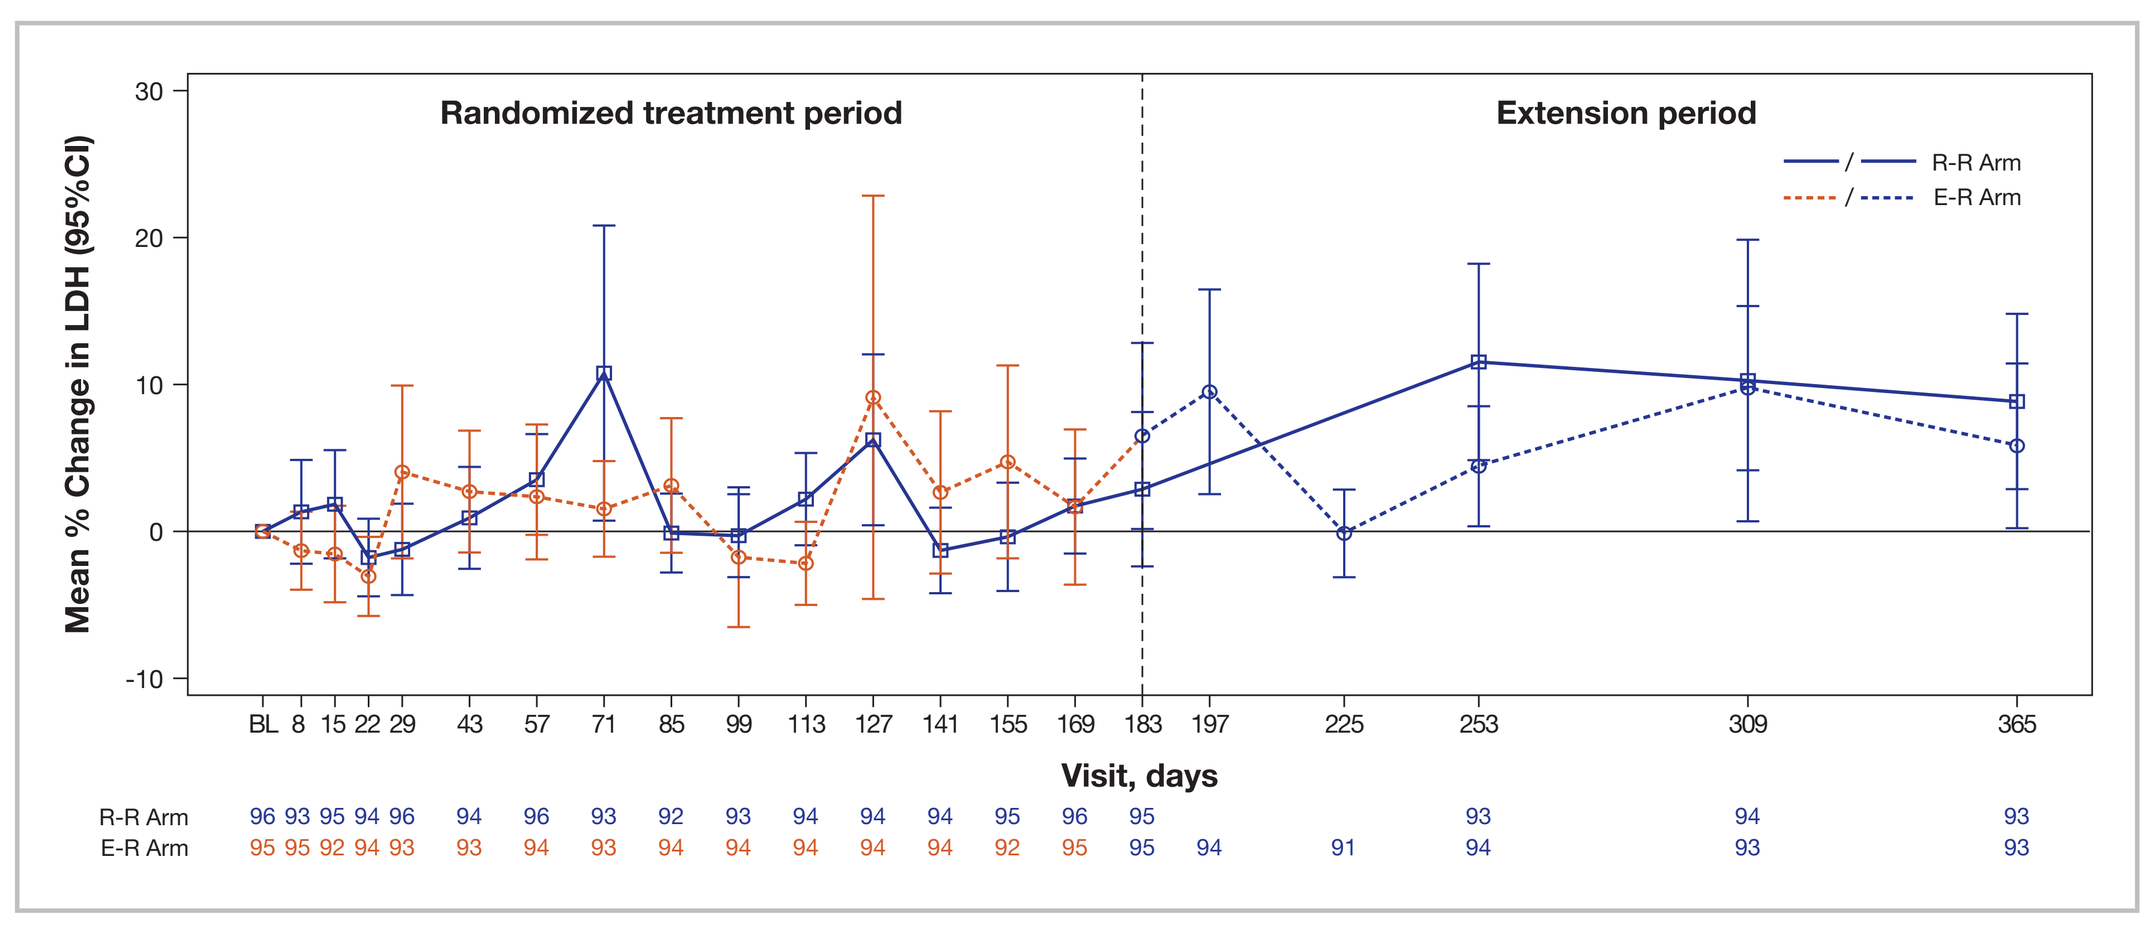

Randomized treatment period
Extension period
R-R Arm
E-R Arm
Mean % Change in LDH (95%CI)
Visit, days

BL, baseline; LDH, lactate dehydrogenase.

Secondary Efficacy Endpoints

Breakthrough Haemolysis

- The proportion of patients who experienced BTH was low and stable over long-term treatment with ravulizumab (Table 1)

Transfusion Avoidance

- In the R-R and E-R arms, 13.4% and 12.2% of patients, respectively, received transfusions within 1 year before receiving their first dose of study treatment
- The percentage of patients avoiding transfusion remained stable in the extension period (weeks 27–52; Table 1)

Hemoglobin Stabilisation

- The percentage of patients with HGB-S in the extension period was consistent with that in the first 26 weeks of the study (Table 1)

Table 1. Summary of Breakthrough Haemolysis Events, Transfusion Avoidance, and Hemoglobin Stabilisation

	R-R Arm (n=97)		E-R Arm (n=98)	
	BL to 26 Weeks[a] (n = 97)	27 to 52 Weeks (n = 96)	BL to 26 Weeks[a] (n = 98)	27 to 52 Weeks (n = 95)
Patients with BTH, n (%)	0	3 (3.1)	5 (5.1)	1 (1.1)
Total number of BTH events/patient (n/n)	0/0	3/3	7/5	1/1
BTH events associated with serum free C5 ≥0.5 µg/mL	0	0	4	0
BTH events associated with infections (with no free C5 elevation)	0	2	2	1[b]
BTH events without elevated free C5 or reported infection	0	1	1	0
Transfusion avoidance, n (%)	85 (87.6)	83 (86.5)	81 (82.7)	79 (83.2)
Hemoglobin stabilization, n (%)	74 (76.3)	78 (81.2)	74 (75.5)	77 (81.1)

BL, baseline; BTH, breakthrough hemolysis.

[a] Data from BL to 26 weeks are based on the full-analysis set.

[b] This patient was 1 of the 2 patients who experienced BTH events with infections (with no free C5 elevation) from BL to 26 weeks.

FACIT-Fatigue

- FACIT-Fatigue scores were maintained in both treatment groups through 52 weeks (Figure 4)

Figure 4. Mean (95% CI) FACIT-Fatigue Scores Over Time

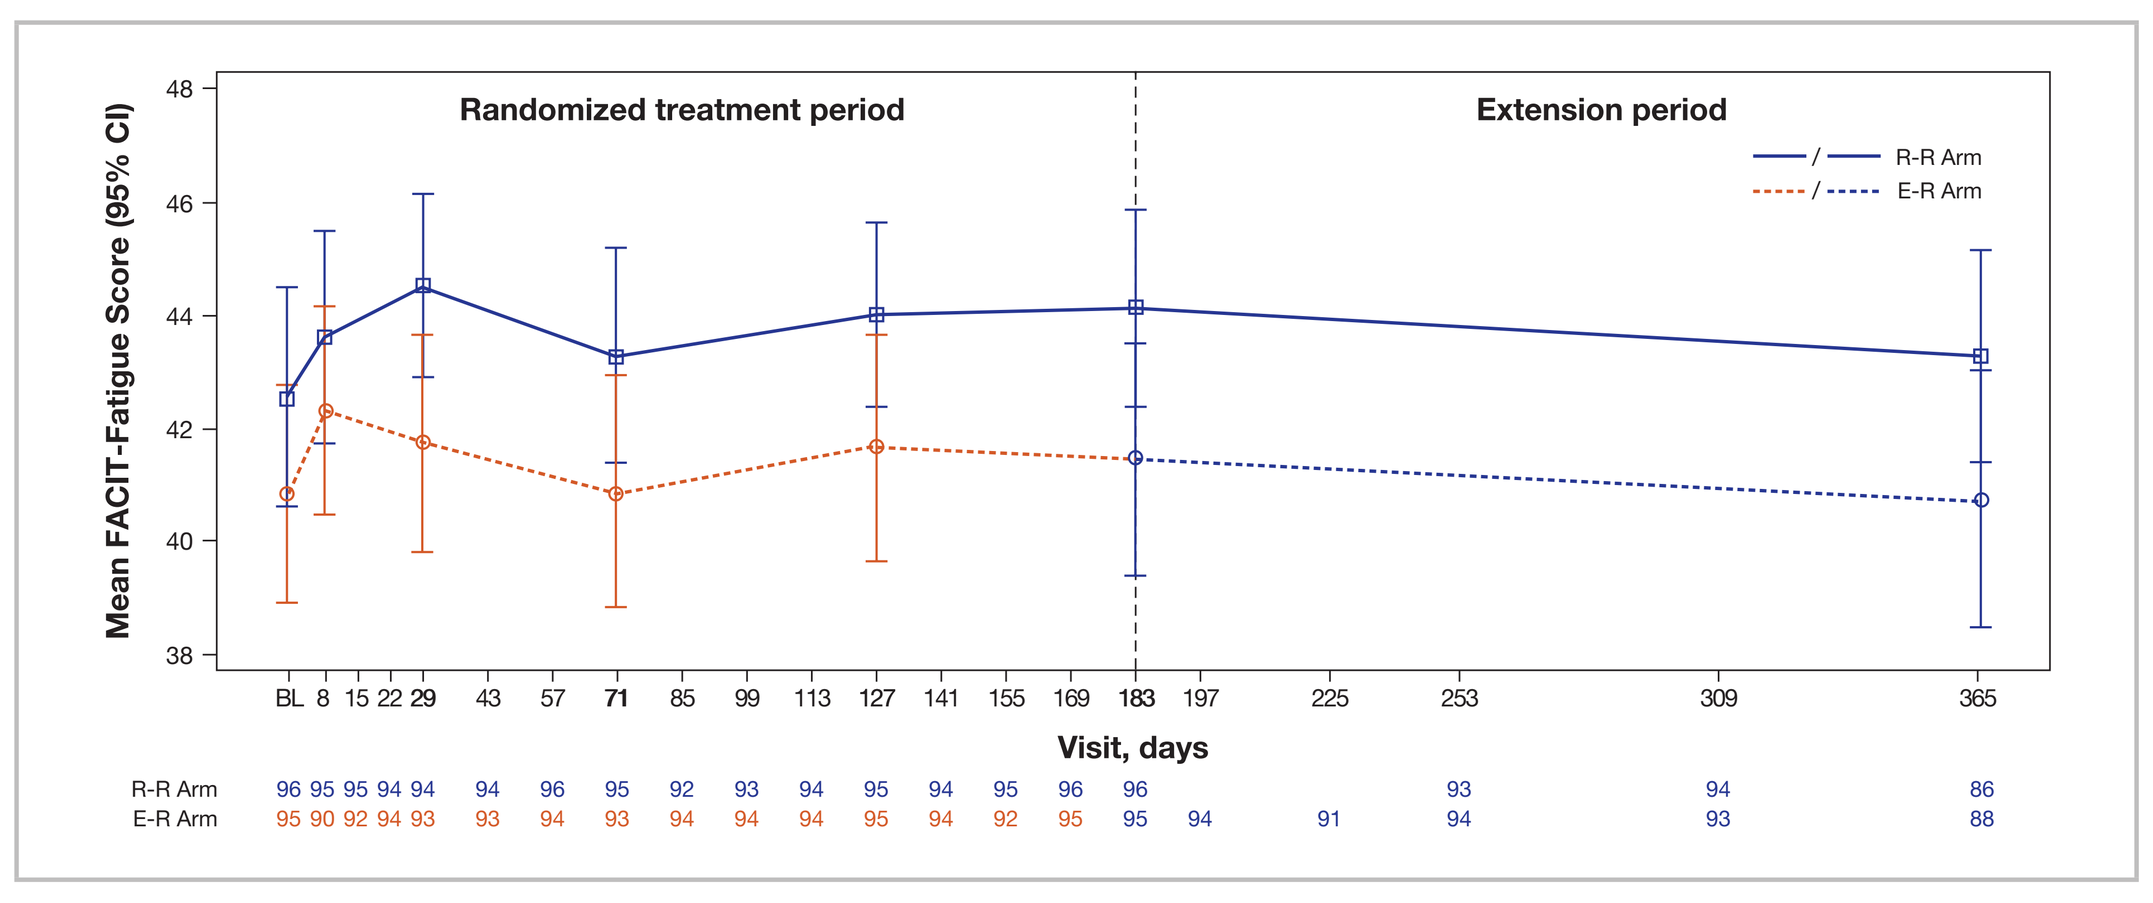

Randomized treatment period
Extension period
R-R Arm
E-R Arm
Mean FACIT-Fatigue Score (95% CI)
Visit, days

BL, baseline.

Other Endpoints

Serum Free C5 Control

- All patients in the R-R arm continued to maintain free C5 < 0.5 µg/mL at all time points through 52 weeks (n = 96; Figure 5)
- Patients in the E-R arm showed improved free C5 control and no patients had free C5 ≥ 0.5 µg/mL following switch to ravulizumab

Figure 5. Box Plotsa of Serum Free C5 Over Time

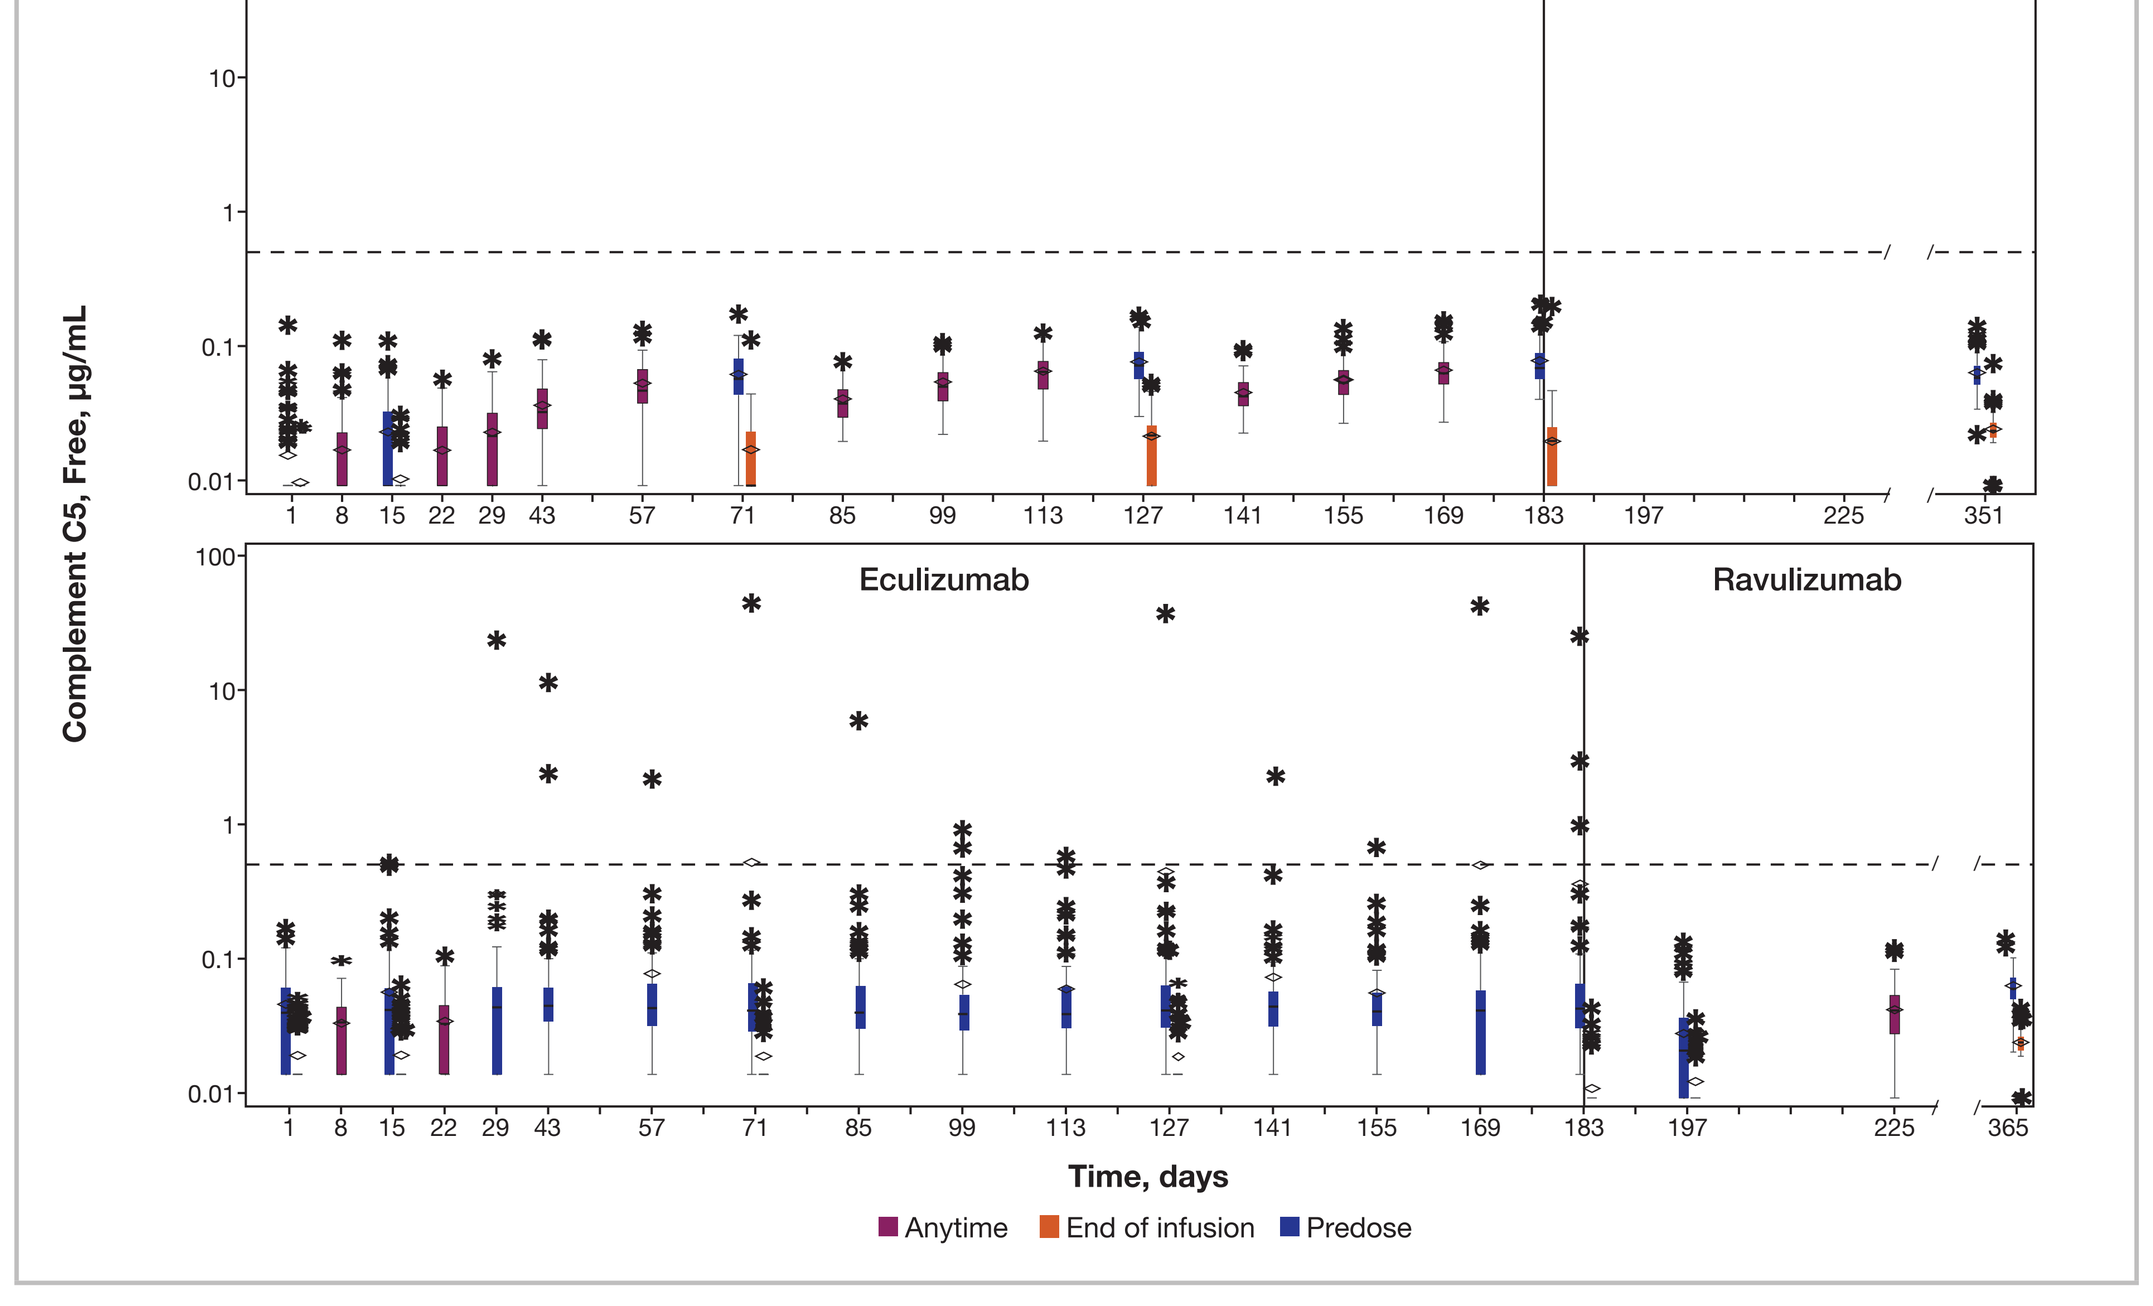

Ravulizumab
Ravulizumab
Eculizumab
Ravulizumab
Complement C5, Free, µg/mL
Time, days
Anytime
End of infusion
Predose

[a] The median is indicated by a horizontal line in the middle of each box. The mean is indicated by a diamond. The 75th and 25th percentiles (interquartile range) are indicated by the top and the bottom borders of the box, respectively. The whiskers represent the 1.5 interquartile range of the lower and upper quartiles, respectively. Outliers are represented by asterisks beyond the whiskers.

Safety

- During weeks 27–52, fatigue was the most common adverse event (incidence ≥10%) in the R-R arm (13 patients [13.5%]), and fatigue (13 [13.7%]) and headache (10 [10.5%]) were most common in the E-R arm (**Table 2**)
- No new treatment-emergent antidrug antibody–positive response was reported during weeks 27–52
- There were no meningococcal infections, deaths, or discontinuations due to adverse events

Table 2. Adverse Events Summary

Variables	R-R Arm (n=97)		E-R Arm (n=98)	
	BL to 26 Weeks (n = 97)	27 to 52 Weeks (n = 96)	BL to 26 Weeks (n = 98)	27 to 52 Weeks (n = 95)
Any AE, n (%)	89 (91.8)	76 (79.2)	86 (87.8)	71 (74.7)
AE leading to study drug dis-continuation, n (%)	0	0	0	0
Any SAE, n (%)	4 (4.1)	8 (8.3)	8 (8.2)	5 (5.3)
SAE leading to study drug discontinuation, n (%)	0	0	0	0
Death, n (%)	0	0	0	0
Most common AEs (≥5% incidence during weeks 27–52), n (%)				
Fatigue	7 (7.2)	13 (13.5)	7 (7.1)	13 (13.7)
URTI	18 (18.6)	9 (9.4)	11 (11.2)	8 (8.4)
Diarrhoea	9 (9.3)	6 (6.3)	7 (7.1)	5 (5.3)
Headache	27 (27.8)	6 (6.3)	19 (19.4)	10 (10.5)
Nasopharyngitis	21 (21.6)	6 (6.3)	20 (20.4)	7 (7.4)
Pyrexia	9 (9.3)	6 (6.3)	5 (5.1)	6 (6.3)
Pain in extremity	5 (5.2)	4 (4.2)	3 (3.1)	5 (5.3)
Dizziness	3 (3.1)	2 (2.1)	7 (7.1)	6 (6.3)
Anaemia	6 (6.2)	1 (1.0)	3 (3.1)	5 (5.3)
Back pain	4 (4.1)	1 (1.0)	4 (4.1)	6 (6.3)

AE, adverse event; BL, baseline; SAE, serious adverse event; URTI, upper respiratory tract infection.

REFERENCES

1. ULTOMIRIS (ravulizumab) [package insert]. Boston, MA: Alexion Pharmaceuticals, Inc; 2019.
2. Ultomiris (ravulizumab) [summary of product characteristics]. Levallois-Perret, France: Alexion Europe SAS; 2019.
3. ULTOMIRIS® (ravulizumab) Receives Marketing Authorization From Japan's Ministry Of Health, Labour And Welfare (MHLW) For The Treatment Of Adults With Paroxysmal Nocturnal Hemoglobinuria (PNH) [press release]. Boston, MA: Alexion Pharmaceuticals; June 18, 2019. https://news.alexionpharma.com/press-release/product-news/ultomiris-ravulizumab receives-marketing-authorization-japans-ministry-he. Accessed August 29, 2019.
4. Government of Canada. Regulatory decision summary – Ultomiris – Health Canada. https://hpr-rps.hres.ca/reg-content/regulatory-decision-summary-detail.php?lang=en&linkID=RDS00547. Accessed September 17, 2019.
5. Agência Nacional de Vigilância Sanitária. Registro ANVISA nº 1981100040011 - Ultomiris. https://www.smerp.com.br/anvisa/?ac=prodDetail&anvisaId=1981100040011. Accessed September 17, 2019.
6. Government of Australia. Prescription Medicine Decision Summary - Ultomiris. https://www.tga.gov.au/apm-summary/ultomiris. Accessed December 3, 2019.
7. Kulasekararaj AG, et al. *Blood*. 2019;133(6):540–549.

ACKNOWLEDGMENTS

The investigators and sponsor thank the patients and their families for their participation in and support for this clinical study. The authors would like to acknowledge Clinical Thinking, Inc. (Hamilton, NJ, USA) which provided editorial and medical writing support by Apurva Davé, PhD, with funding from Alexion Pharmaceuticals, Inc. (Boston, MA, USA).

DISCLOSURES

A. Kulasekararaj Conflict with: Honoraria from Alexion Pharmaceuticals, Inc., Amgen, Celgene, Novartis, and Ra Pharma; Board of Directors or advisory board member for Alexion Pharmaceuticals, Inc., Amgen, Celgene, Novartis, and Ra Pharma; and consulting fees from Achilleon, Akari Therapeutics, Alexion Pharmaceuticals, Inc., AmCelgene, and Novartis., L. Mitchell Conflict with: Honoraria from Alexion Pharmaceuticals, Inc., A. Hill Conflict with: Honoraria and/or consultancy from Akari Therapeutics, Alexion Pharmaceuticals, Inc., Apellis, Bioverativ, Novartis, Ra Pharma, Regeneron, and Roche, S. Langemeijer Conflict with: Honoraria and consulting fees from Alexion Pharmaceuticals, Inc., R. Wells Conflict with: Honoraria and research funding from Alexion Pharmaceuticals, Inc., Celgene, and Novartis and consulting fees from Alexion Pharmaceuticals, Inc. , F. A. González Fernández Conflict with: Honoraria, consulting/speakers bureau fees, and research funding from Alexion Pharmaceuticals, Inc., A. Gaya Conflict with: Honoraria and consulting fees from Alexion Pharmaceuticals, Inc., Novartis, and Merck Sharpe & Dohme and Board of Directors or advisory board member for Alexion Pharmaceuticals, Inc. , E. O. Gutiérrez Conflict with: Honoraria and consulting fees from Alexion Pharmaceuticals, Inc., C. Piatek Conflict with: Honoraria from and Board of Directors or advisory board member for Alexion Pharmaceuticals, Inc., Dova, and Rigel; research funding from Alexion Pharmaceuticals, Inc., and Incyte; and Speakers Bureau for Dova., K. Usuki Conflict with: Honoraria and research funding from Alexion Pharmaceuticals, Inc., Kyowa-Kirin, and Novartis and research funding from Chugai Pharmaceutical., A. Bosi: None Declared, R. Brodsky Conflict with: Research funding from Achillion and Board of Directors or advisory board member for and grant funding from Alexion Pharmaceuticals, Inc., M. Ogawa Conflict with: Employee and stockholder of Alexion Pharmaceuticals, Inc., J. Yu Conflict with: Employee and stockholder of Alexion Pharmaceuticals, Inc., S. Ortiz Conflict with: Employee and stockholder of Alexion Pharmaceuticals, Inc., A. Röth Conflict with: Honoraria from Alexion Pharmaceuticals, Inc., Bioverativ, Novartis, and Roche; consulting fees from Alexion Pharmaceuticals, Inc., Apellis, Bioverativ, Novartis, and Roche; Board of Directors or advisory board member for Alexion Pharmaceuticals, Inc., Apellis, Novartis, and Roche; and research funding from Roche., J. W. Lee Conflict with: Honoraria, consulting fees, and research support (to Seoul St. Mary's Hospital) from Alexion Pharmaceuticals, Inc.; Board of Directors or advisory board member for Alexion Pharmaceuticals, Inc.; and research funding from Achillion., R. Peffault de la Tour Conflict with: Consultancies, honoraria, and research funding from Alexion Pharmaceuticals, Inc., Novartis, and Pfizer and research funding from Amgen.

Encore presented at BSH, virtual conference, November 2020. Originally presented at American Society of Hematology (ASH) 61st Annual Meeting, December 2019

Alexion UK has provided agency support in the development of this poster.

BSH2020 PO-146-A

Red Cell Disorders

Colin Griffin

Poster presented at: British Society for Haematology Listening • Learning • Leading

PosterSessionOnline.com