Do we underestimate the physical and emotional impact of early stage ADPKD? Evidence for a discrepancy between patient experience and physician perceptions

James Marsh[1], Anna Baker[2], Andrew Makin[3], Alison Carr[4]

[1]St. Helier Hospital, Sutton, UK; [2]London Metropolitan University, London, UK; [3]Otsuka Europe Development and Commercialisation, Buckinghamshire, UK; [4]Hamell, London, UK

BACKGROUND

- Autosomal dominant polycystic kidney disease (ADPKD) is the most common inherited renal disorder[1]
- The early stages of ADPKD are often described as asymptomatic[2]
- However, emerging evidence has reported that patients can suffer from pain and fatigue during these early stages
- There is also the possibility that early stage ADPKD can have a substantial psychological effect on patients

OBJECTIVES

1) To explore the self-reported physical and emotional impact of early stage ADPKD
2) To determine physicians' perceived impact of the early stages of the disease

METHODS

Study design

- Cross sectional study in the following countries:

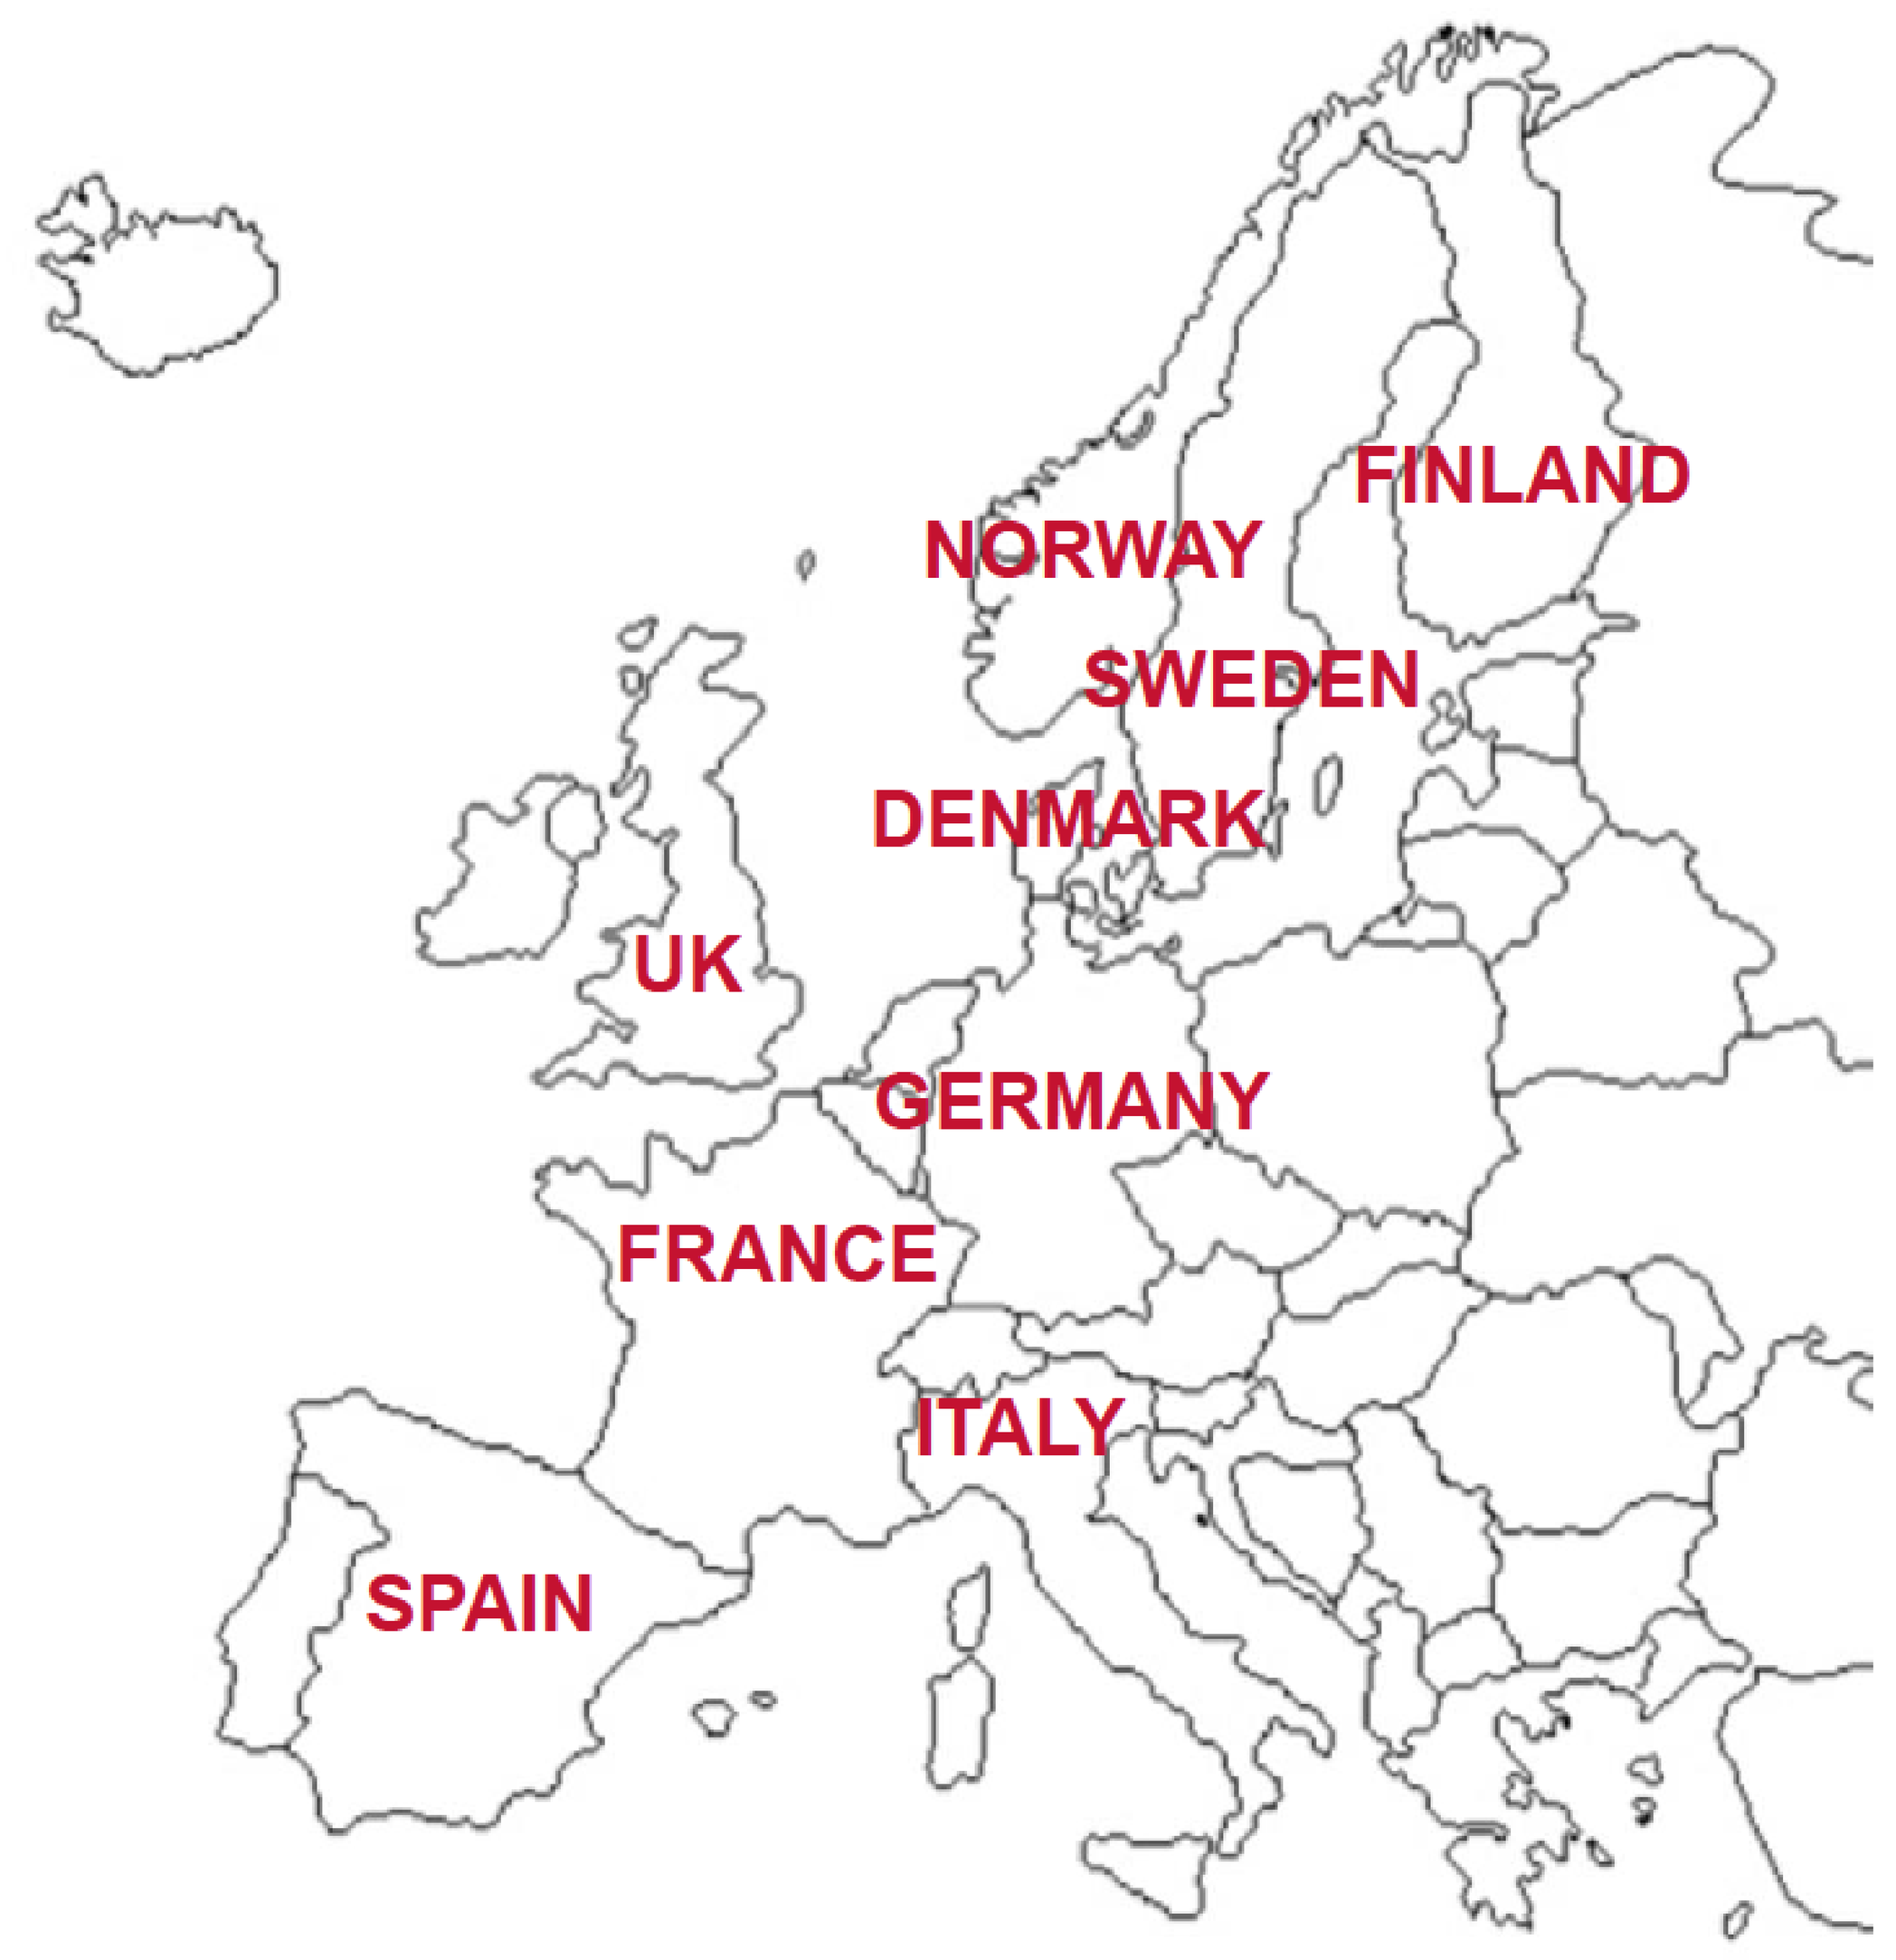
FINLAND
NORWAY
SWEDEN
DENMARK
UK
GERMANY
FRANCE
ITALY
SPAIN

Subjects

- 80 adult (≥18 years) patients with a diagnosis of ADPKD 757 primary care physicians (PCPs) and consultant nephrologists

Data collection

- All 80 patients, 72 nephrologists and 85 PCPs participated in semi-structured interviews
- Reponses were used to construct online questionnaires that were completed by 300 nephrologists and 300 PCPs

Analysis

- Interview transcripts were analysed using thematic analysis
- Each interview was independently read, coded and commented on by two qualitative researchers

OPEL/0514/TOL/1077 20.05.14

RESULTS

Patient experiences

Physical symptoms

- 31% patients had early (CKD stage 1 or 2) ADPKD
- Only 3 patients were completely asymptomatic. All the others reported physical symptoms in early stage disease
- Physical symptoms reported included pain, fatigue, breathlessness, weakness and general malaise
- These symptoms were severe enough to inhibit work and physical activity/exercise

Emotional impact

- The emotional impact was widespread, caused many patients to revaluate their life expectations, and was represented by 3 main themes:

1. Loss
- In relation to having children (inherited disease), life expectancy, healthy living, and valued lifestyle activities

2. Uncertainty
- About the progression of the disease, what their experience would be of the process and when it would happen

3. Fear
- For themselves and their children, exacerbated by uncertainty

- Patients often reported that the information provided by physicians was unhelpful
- Patients with early stage disease often based their expectations on the experience of their affected family members and believed their disease would follow the same course
- The words patients remembered physicians saying at diagnosis were:

'Fatal'

'Worse and worse'

'Incurable'

Physician experiences

- **Tables 1 and 2** report the physician perceived impact of early stage ADPKD
- 24% nephrologists and 16% of PCPs thought that patients did not experience **any** physical symptoms
- 67% of nephrologists and 65% of PCPs thought that patients experienced only **mild** physical symptoms
- There was a greater recognition for the emotional impact of early stage ADPKD

Table 1: Physicians' perception of the severity of **physical symptoms** experienced by patients during early stage ADPKD, by country and physician type.

	France %		Germany %		Italy %		Nordic %		Spain %		UK %		Total %	
	NP	PCP	NP	PCP	NP	PCP	NP	PCP	NP	PCP	NP	PCP	NP	PCP
None	26	28	24	18	16	12	28	17	36	2	16	20	24	16
Mild	63	56	64	54	72	66	68	81	62	63	72	73	67	65
Moderate	12	16	10	26	12	22	4	2	2	33	12	8	9	18
Severe	0	0	2	2	0	0	0	0	0	2	0	0	0	1
Total	100	100	100	100	100	100	100	100	100	100	100	100	100	100

KEY: NP=Nephrologists; PCP=primary care physicians; N=sample size

Table 1: Physicians' perception of the severity of **emotional symptoms** experienced by patients during early stage ADPKD, by country and physician type

	France %		Germany %		Italy %		Nordic %		Spain %		UK %		Total %	
	NP	PCP	NP	PCP	NP	PCP	NP	PCP	NP	PCP	NP	PCP	NP	PCP
None	4	10	10	8	2	0	4	15	8	2	2	8	5	7
Mild	53	41	46	42	16	34	68	38	48	57	50	59	47	45
Moderate	33	45	34	44	66	62	26	38	34	39	46	31	40	43
Severe	10	4	10	6	16	4	2	10	10	2	2	2	8	5
Total	100	100	100	100	100	100	100	100	100	100	100	100	100	100

KEY: NP=Nephrologists; PCP=primary care physicians; N=sample size

CONCLUSIONS

- The emotional and physical impact of early stage ADPKD is widely reported by patients, however, the magnitude of this impact, particularly the physical impact, is underestimated by physicians
- Physicians were more likely to acknowledge physical symptoms if there was a plausible medical explanation (e.g. kidneys that were large enough to cause distention)
- This under recognition may lead to physicians ignoring or discounting symptoms, leading to further distress and a reduction in quality of life for patients
- Further support is required for patients at the point of diagnosis of ADPKD

REFERENCES

(1) Masoumi *et al.* Ther clin risk manag. 2008; 4(2): 393-407. (2) Rizk *et al.* Ped Neph. 2008;23(7):1029-1036. (3)Miskulin *et al.* Am J Kid Dis. 2014;63(2):214-226. (4) Bajwa *et al.* Kidney Int. 2004; 66(4):151-1569.

ACKNOWLEDGEMENTS

Thank you to all patients and physicians who took part in the study, the patient organisations in the participating countries and the international ADPKD patient organisation.

51ERA-EDTA 20--SP

C) Renal development and cystic diseases.

Alison Carr

DOI: 10.3252/pso.eu.51era.2014

Poster presented at: 51 ERA-EDTA Congress Amsterdam 2014

PosterSessionOnline.com