The effect of dietary phosphate on FGF-23 secretion in patients with diabetic and non-diabetic moderate to advanced chronic kidney disease

Katarzyna Muras, Anna Masajtis-Zagajewska, Michał Nowicki
Department of Nephrology, Hypertension and Kidney Transplantation
Medical University of Lodz, Poland

MEDICAL UNIVERSITY OF LODZ

Background

Increased serum concentration of fibroblast growth factor 23 (FGF-23) has been linked to worse prognosis in chronic kidney disease (CKD). It has been recently shown that the responses of both FGF-23 and parathormone (PTH) to an increased oral phosphate intake are impaired in patients with early CKD and type 2 diabetes mellitus (DM).

Aim

The assessment of the effect of oral phosphate intake on serum FGF-23 and other parameters of calcium-phosphate metabolism in diabetic and non-diabetic CKD patients with moderate to severe renal function impairment.

Subjects and methods

- ✓ **The study comprised 26 patients with CKD; 15 (6F, 9M) with diabetic and 11 (3F, 8M) with non-diabetic nephropathy**
- ✓ **All patients received 7-day high-phosphate diet (1000 mg/24h) with additional intake of 90 mL of sodium and potassium phosphate as a solution containing: Na_2HPO_4/K_2HPO_4 and NaH_2PO_4/KH_2PO_4 in 4:1 ratio (31 mg of phosphate, 0.9 mEq sodium and 0.9 mEq potassium per 5 mL)**
- ✓ **At baseline and after 3 and 7 days of high phosphate diet serum concentration of FGF-23, Ca, P and 1,25(OH)$_2$D was measured and the urine was collected for Ca and PO_4**

Clinical and biochemical parameters at baseline

	Non-diabetic patients	Diabetic patients	All patients	p-value
Male/Female	8/3	9/6	17/9	p>0.05
Age (yr)	58.3±17.8	67.9±8.3	63.8±13.7	p=0.11
BMI (kg/m^2)	27.5±5.0	26.9±3.9	27.1±4.3	p>0.05
eGFR$_{C-G}$ (mL/min/1.73 m^2)	32.8±24.3	30.3±12.9	31.4±18.2	p=0.76
Ca (mmol/L)	2.22±0.16	2.20±0.11	2.2±0.13	p=0.84
PO_4 (mmol/L)	1.15±0.21	1.28±0.23	1.2±0.2	p=0.15
PTH (pg/mL)	223.0±192.0	150.0±77.0	179.0±135.0	p=0.3
25 OH vitamin D (pg/mL)	5.32±3.99	4.58±2.78	4.89±3.28	p=0.6
1,25 (OH)$_2$ vitamin D (pg/mL)	12.2±7.8	11.8±7.4	11.9±7.4	p=0.9
FGF-23 (pg/mL)	75.0±84.3	58.0±52.7	65.1±66.5	p=0.58
MCP-1 (pg/mL)	63.9±41.8	77.0±47.6	71.5±44.9	p=0.56
Urine – PO_4/Cr (dmol/g)	0.10±0.05	0.16±0.07	0.14±0.07	p=0.012
Urine - Ca/Cr (dmol/g)	0.008±0.006	0.012±0.011	0.010±0.009	p=0.25

Results

In diabetic CKD patients serum calcium decreased from 2.21±0.11 mmol/L at baseline to 2,12±0.14 and 2.08±0.23 mmol/L on day 3 and 7, respectively (p<0.01). The change of serum calcium was not significant in non-DM CKD patients. Serum phosphorus was 1.28±0.24, 1.41±0.26 and 1.44±0.34 mmol/L, respectively at baseline and on day 3 and 7 of the study in DM group and 1.15±0.21, 1.31±0.22 and 1.46±0.40 mmol/L, respectively in non-DM (p<0.01). Serum PTH increased from 179±135 to 279±252 pg/mL on day 7 in all patients (p=0.01) and the change of PTH was similar in DM and non-DM patients. 1,25(OH)$_2$D levels did not change during the study and were 11.9±7.4, 11.7±8.24, 12.8±8.9 pg/mL, respectively in the whole study population, and similar in DM and non-DM subjects. Serum FGF-23 in DM patients increased from 58.1±52.7 at baseline to 91.6±71.1 pg/mL on day 3 (p=0.001) but latter tended to decrease (6.3±52.4 pg/mL on day 7). In non-DM patients there was a steady increase of FGF-23 from 75±84.3 at baseline to 102.1±103.6 on day 3 and 176±197 pg/mL on day 7 (p=0.04 vs baseline). Urine calcium excretion remained unchanged but phosphate excretion increased significantly to a similar extent in both groups.

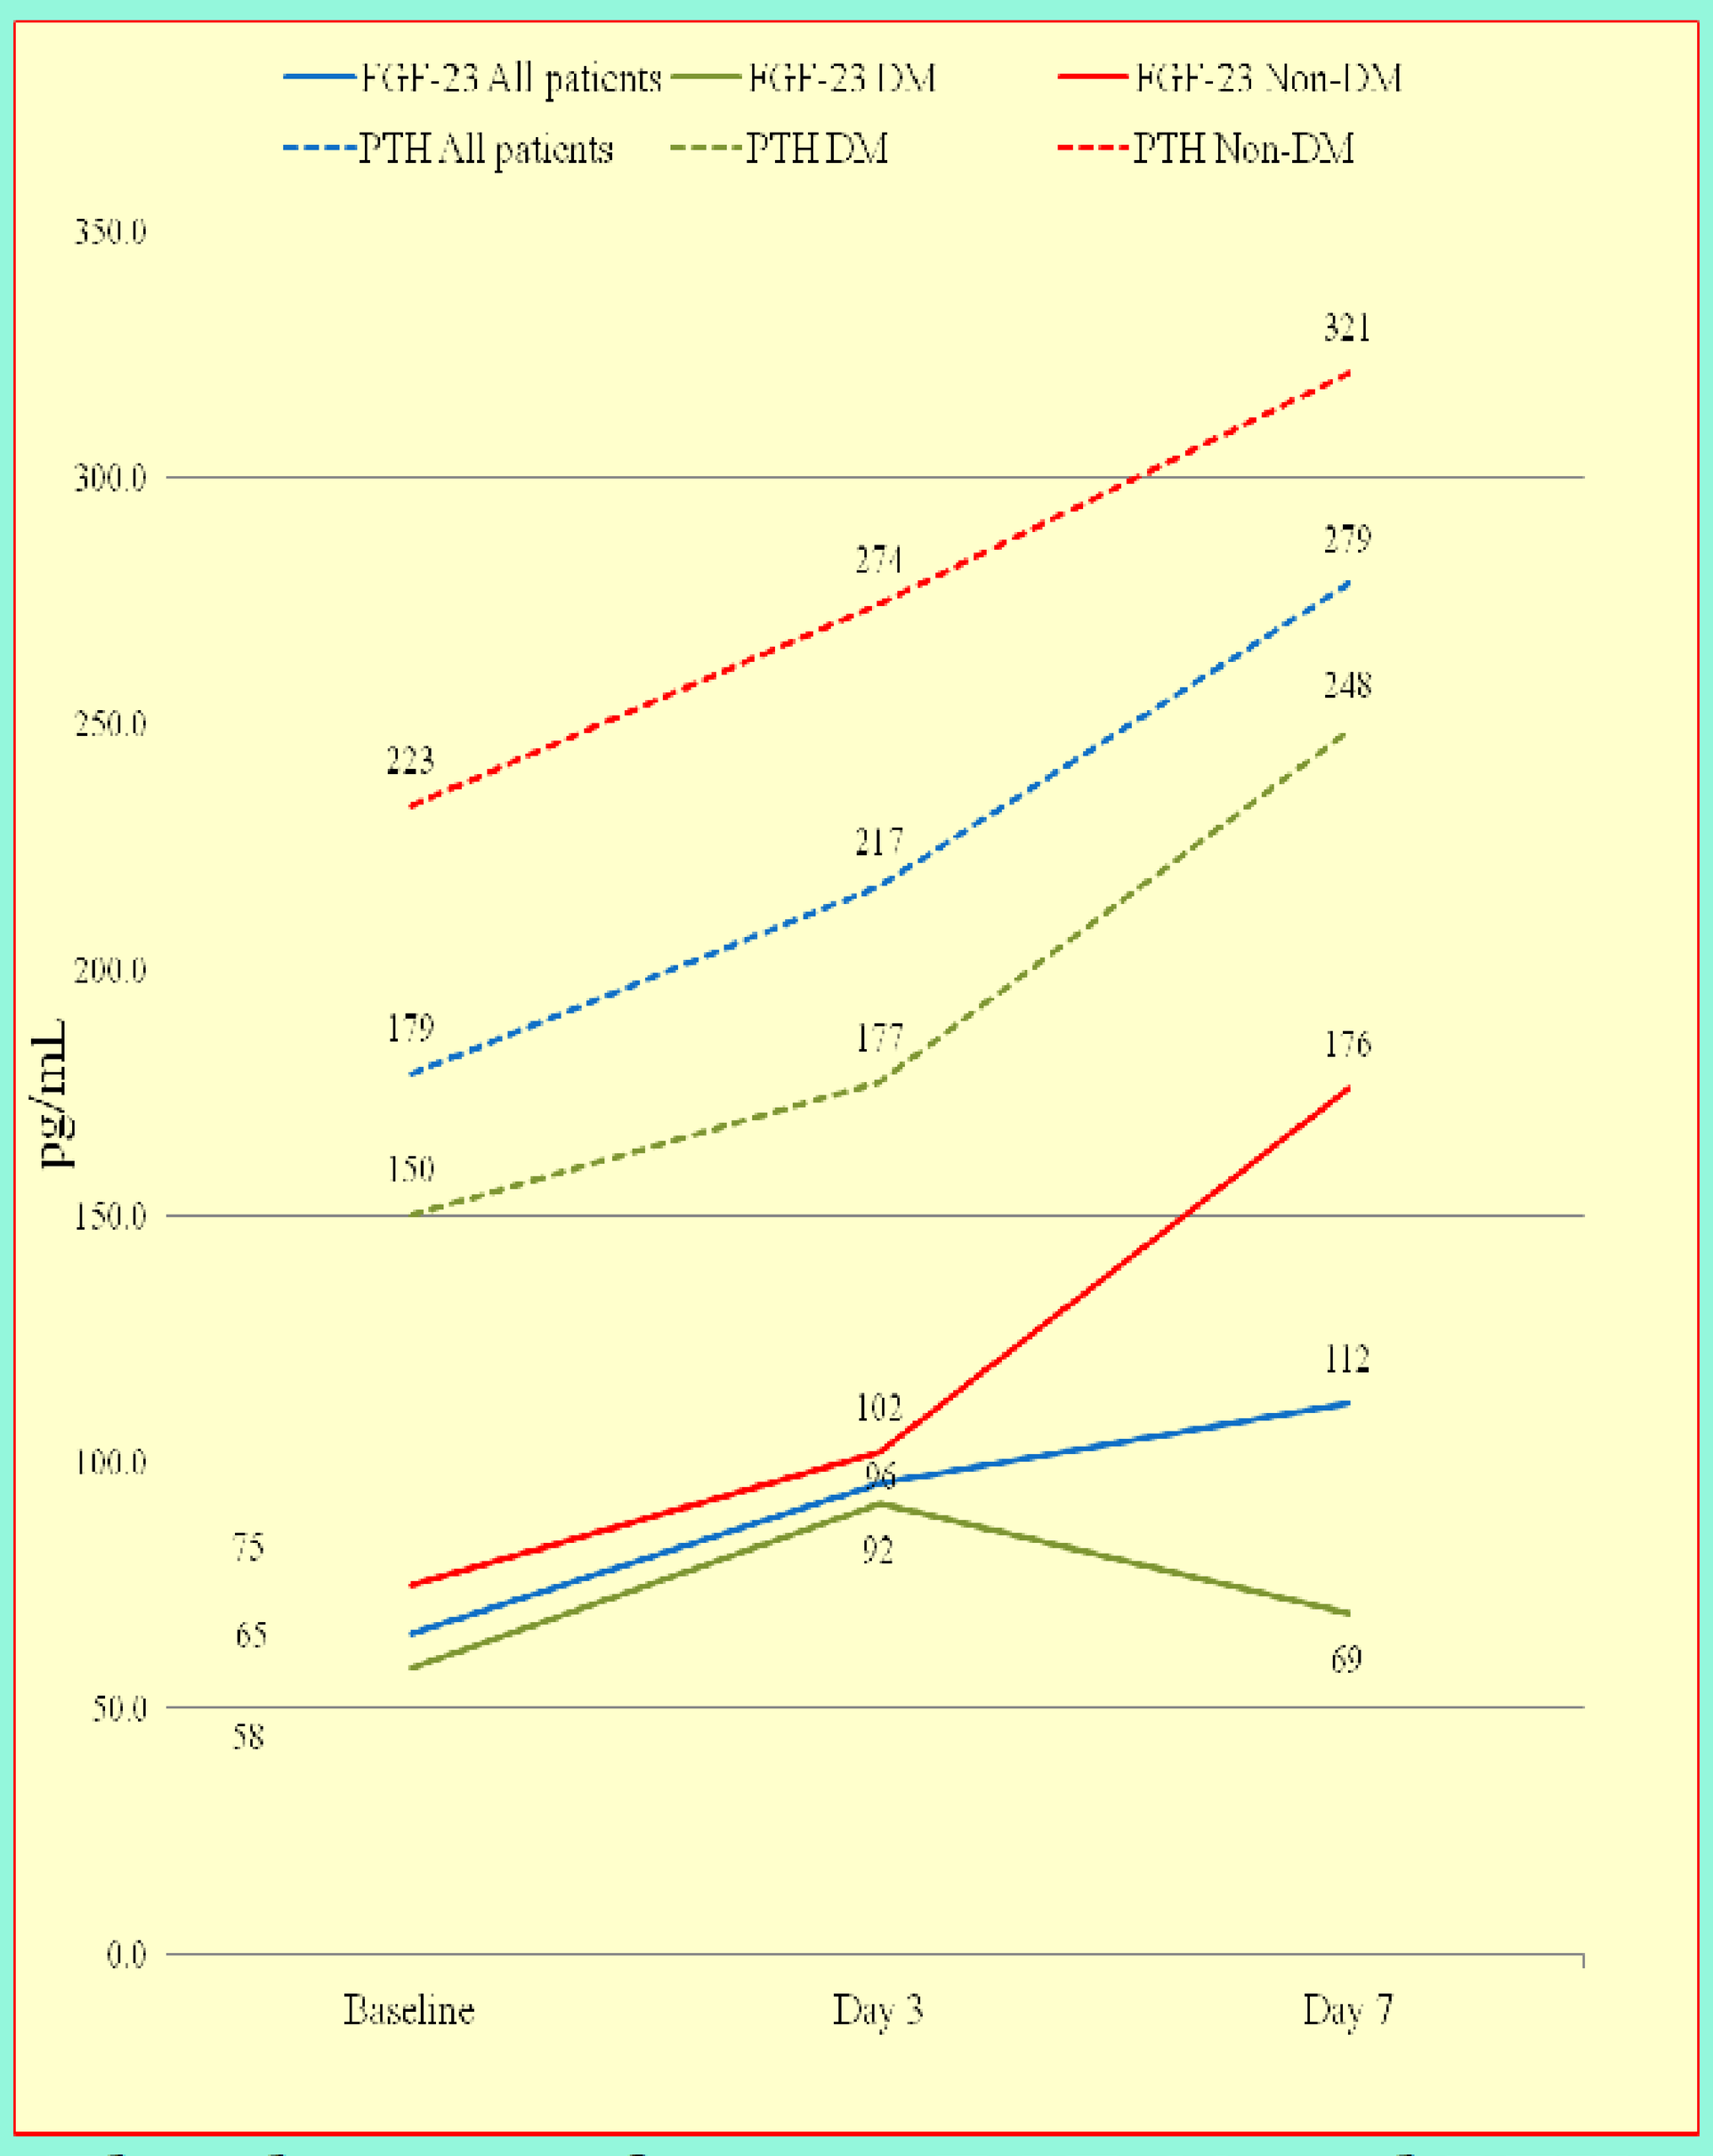
FGF-23 All patients
FGF-23 DM
FGF-23 Non-DM
PTH All patients
PTH DM
PTH Non-DM
pg/mL
350.0
300.0
250.0
200.0
150.0
100.0
50.0
0.0
Baseline
Day 3
Day 7
321
279
274
248
223
217
179
177
176
150
112
102
96
92
75
69
65
58

The changes of serum FGF-23 and PTH caused by high-phosphate intake

Conclusions

Renal phosphate excretion in response to high oral phosphate intake is preserved in diabetic patients with impaired kidney function despite the impaired FGF-23 response. Our results may question the dominant role of FGF-23 in phosphate disposal in moderate to severe diabetic CKD.

50ERA-EDTA
221--MP
Pathophysiology and clinical studies in CKD 1-5.
Katarzyna Muras
Poster presented at: ERA-EDTA 50th Congress Istanbul, Turkey May 18-21, 2013
PosterSessionOnline.com