Contrasting approaches to predicting ADPKD progression and outcomes

O'Reilly R[1], McEwan P[2,3], Bennett H[2], Robinson R[1]

[1] Otsuka Pharmaceutical Europe Ltd, Wexham, UK; [2] Health Economics & Outcomes Research Ltd, Monmouth, UK;
[3] School of Human & Health Sciences, Swansea University, Swansea, UK

INTRODUCTION AND AIMS

Autosomal dominant polycystic kidney disease (ADPKD) is a major cause of end-stage renal disease (ESRD) [1], characterised by progressive development of kidney cysts leading to complications such as pain, haematuria and hypertension, and declining renal function.

Given the heterogeneous nature of progression, an understanding of a patient's risk of, and likely rate of progression towards, ESRD is important to inform treatment decisions.

Rates of ADPKD progression are linked to factors such as age, gender, genetic mutation and hypertension, in addition to total kidney volume (TKV) [2,3].

The imaging classification of ADPKD was developed from Mayo clinic observations to inform patient selection for clinical trial inclusion; risk of renal progression is stratified by cyst distribution, age and height-adjusted TKV (classes 1A-E, 2A-B) [4]. The Mayo classification may be used to predict future eGFR for class 1 individuals via a published model [4] and to prioritise patients likely to benefit most from treatment [5].

The ADPKD Outcomes Model (OM) [6] utilises multivariate regression equations incorporating age, gender and TKV derived from the placebo arm of the TEMPO 3:4 trial [7] to predict underlying ADPKD progression; the model can also be used to evaluate the impact of therapeutic intervention on progression to ESRD and to simulate long-term outcomes such as ESRD management and life-expectancy [8].

This study aimed to contrast predictions of ADPKD progression made based on the Mayo classification and using the ADPKD OM, in terms of renal function decline (measured by estimated glomerular filtration rate (eGFR)).

The study also illustrates how predicted renal function trajectories may be utilised to estimate ESRD incidence and consequent requirements for care within the ADPKD OM.

METHODS

Being a population enriched for likelihood of rapid progression, approximately 90% of patients in the TEMPO 3:4 trial were classified as 1C, 1D or 1E at baseline.

Time-dependent renal decline trajectories and time to ESRD (eGFR<15 ml/min/m^2) were predicted utilising the ADPKD OM and Mayo classification for each TEMPO 3:4 subgroup (1C-1E).

Table 1 presents the mean baseline characteristics observed for each subgroup; despite having similar levels of renal function at baseline, there was a trend of higher TKV at a younger age in the mean profiles of 1E versus 1D and 1D versus 1C [9]. Gender distribution was assumed to be equal.

Regression coefficients of the ADPKD OM's natural history progression equations and the Mayo classification model were sampled to account for parameter uncertainty in rates of progression and to present a range of simulated renal function trajectories for each patient profile.

Table 1. Mean baseline characteristics of TEMPO 3:4 patients classified as 1C, 1D and 1E at baseline

Baseline characteristic	TEMPO 1C	TEMPO 1D	TEMPO 1E
Age (years)	41.1	37.8	35.3
eGFR (ml/min/m^2)	82.52	79.99	81.58
TKV (ml)	1,242	1,830	2,255

eGFR: estimated glomerular filtration rate, TKV: total kidney volume
N=1,273; remaining patients classified as 1B (96) or 2 (42)

Table 2. TKV boundaries of Mayo class 1C, 1D and 1E for mean ages observed in each TEMPO 3:4 subgroup

Characteristic	1C	1D	1E
Age (years)	41	37	35
Lower TKV class bound* (ml)	836	1,269	1,914
Upper TKV class bound* (ml)	1,641	2,332	NA

TKV: total kidney volume
* TKV range based on bounds of each Mayo class and height 1.66-1.8m

The impact of within-class variation of TKV was assessed in the ADPKD OM using the age-dependent upper and lower bounds of the Mayo classification (Table 2), while all other characteristics were held constant. Heights of 1.66m (lower) and 1.8m (upper) were utilised based on the TEMPO 3:4 data to convert height-adjusted TKV boundaries of each Mayo class to TKV values.

Finally, long-term outcomes were predicted within the ADPKD OM (parameterised to a UK setting [8]) for cohorts of patients with each TEMPO 3:4 subgroup profile.

The following outcomes were estimated over a lifetime horizon:

- Percentage of patients expected to reach ESRD prior to death
- Number of transplantations conducted
- Number of years on dialysis

RESULTS

Figure 1 presents trajectories of renal function decline (eGFR) for each patient profile predicted using each model.

There was a high degree of overlap between the 95% prediction regions of the Mayo classification and ADPKD OM, with agreement between the approaches highest for the 1E profile and lowest for the 1D profile.

On average, predicted renal decline was more rapid in the ADPKD OM compared to the Mayo classification.

The larger degree of uncertainty presented around predictions made based on the Mayo classification may be influenced by differences in the implemented methods of sampling due to data limitations (i.e. multivariate in ADPKD OM versus independent for Mayo classification).

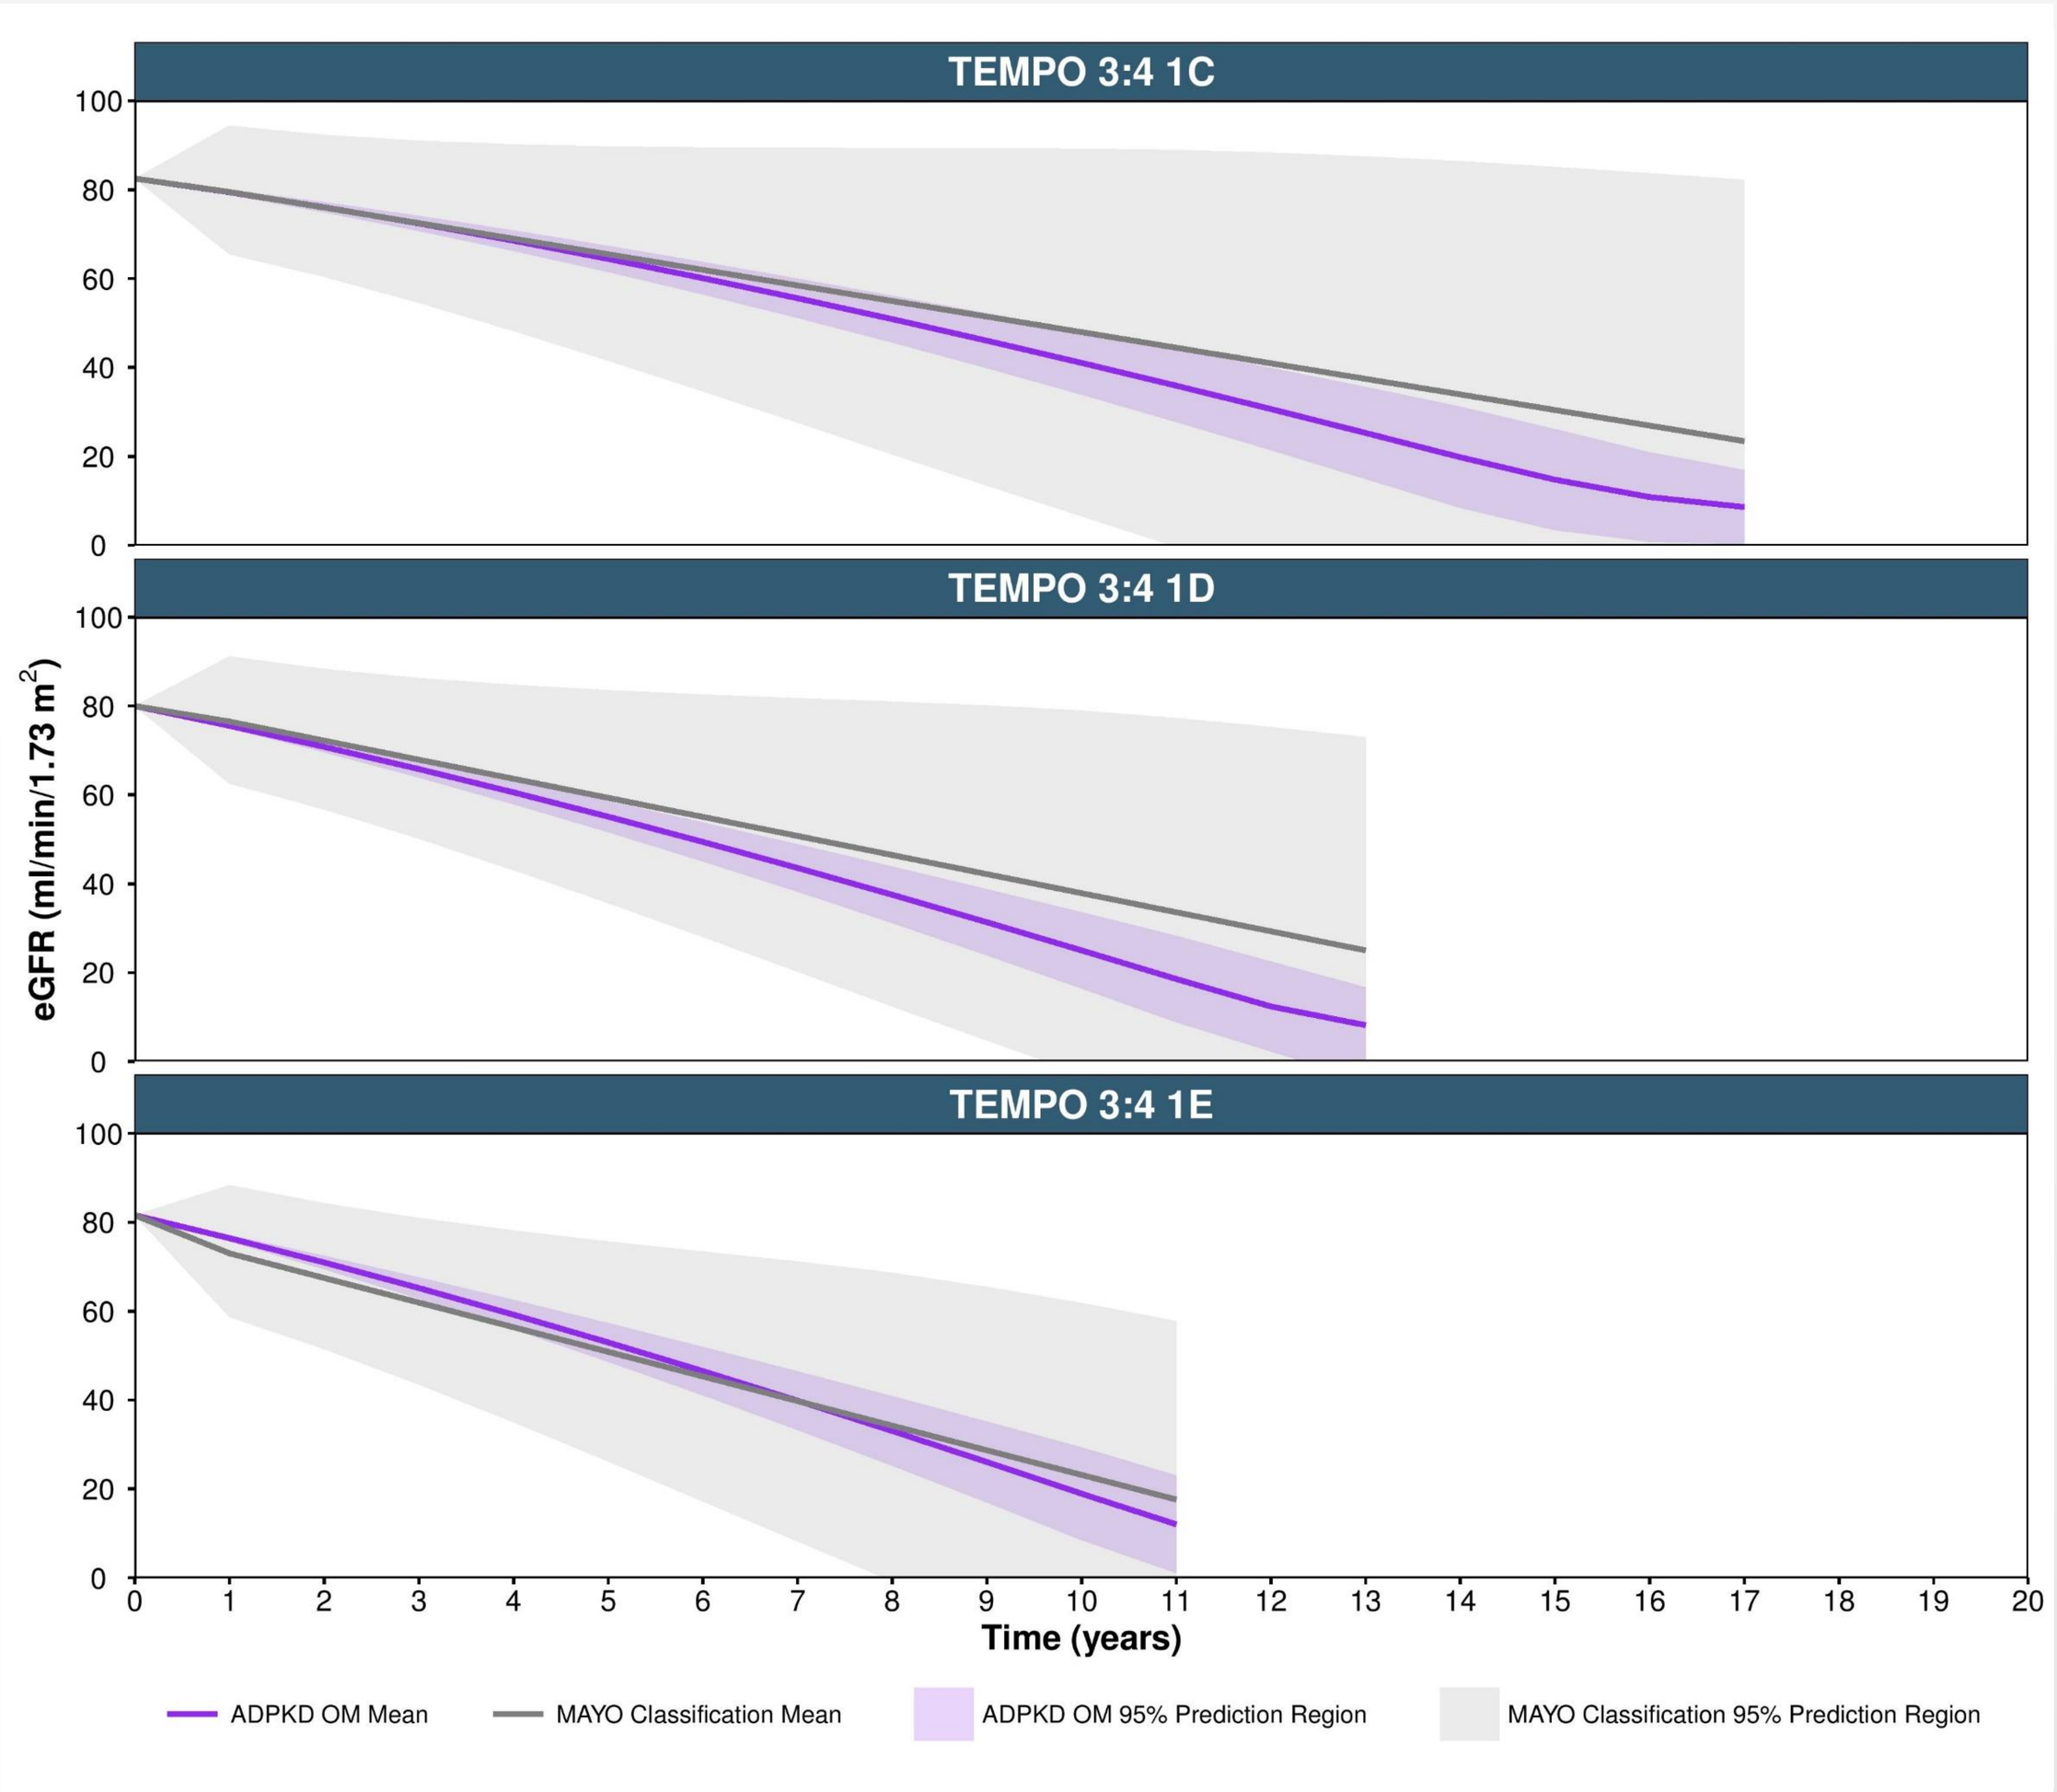

TEMPO 3:4 1C
TEMPO 3:4 1D
TEMPO 3:4 1E
eGFR (ml/min/1.73 m²)
Time (years)
ADPKD OM Mean
MAYO Classification Mean
ADPKD OM 95% Prediction Region
MAYO Classification 95% Prediction Region

Figure 1. Renal decline for TEMPO 3:4 1C, 1D and 1E profiles predicted using the Mayo Classification and ADPKD OM

Table 3. Long-term clinical outcomes predicted for TEMPO 3:4 1C, 1D and 1E profiles within the ADPKD OM

Predicted outcome	TEMPO 1C	TEMPO 1D	TEMPO 1E
Lifetime incidence of ESRD	96.3%	98.3%	98.7%
Mean time to ESRD	15.4	12.1	11.1
Mean years of dialysis	5.0	5.7	5.9
Number of transplants (per 100 individuals)	33	50	54

Under each model, the predicted number of years to the onset of ESRD fell from Mayo Class 1C to 1D and from 1D to 1E, as shown in Figures 1 and 2.

Predicted age at ESRD onset followed the same trend: 56.1 and 60.6 years for the mean 1C profile under ADPKD OM and Mayo classification, respectively, versus 46.3 and 46.8 years for the mean 1E profile.

The largest range of times to ESRD predicted in the ADPKD OM as a consequence of within-class variation in TKV was predicted for the class 1C profile (Figure 2).

For the 1C and 1D profiles, the ADPKD OM predicted ranges of 13 to 20 years and 11 to 15 years, respectively, compared to 19.5 years and 15.5 years using the Mayo classification.

For class 1E, which has no upper TKV bound, the ADPKD OM predicted times to ESRD of up to 12 years, compared to 11.5 years using the Mayo classification.

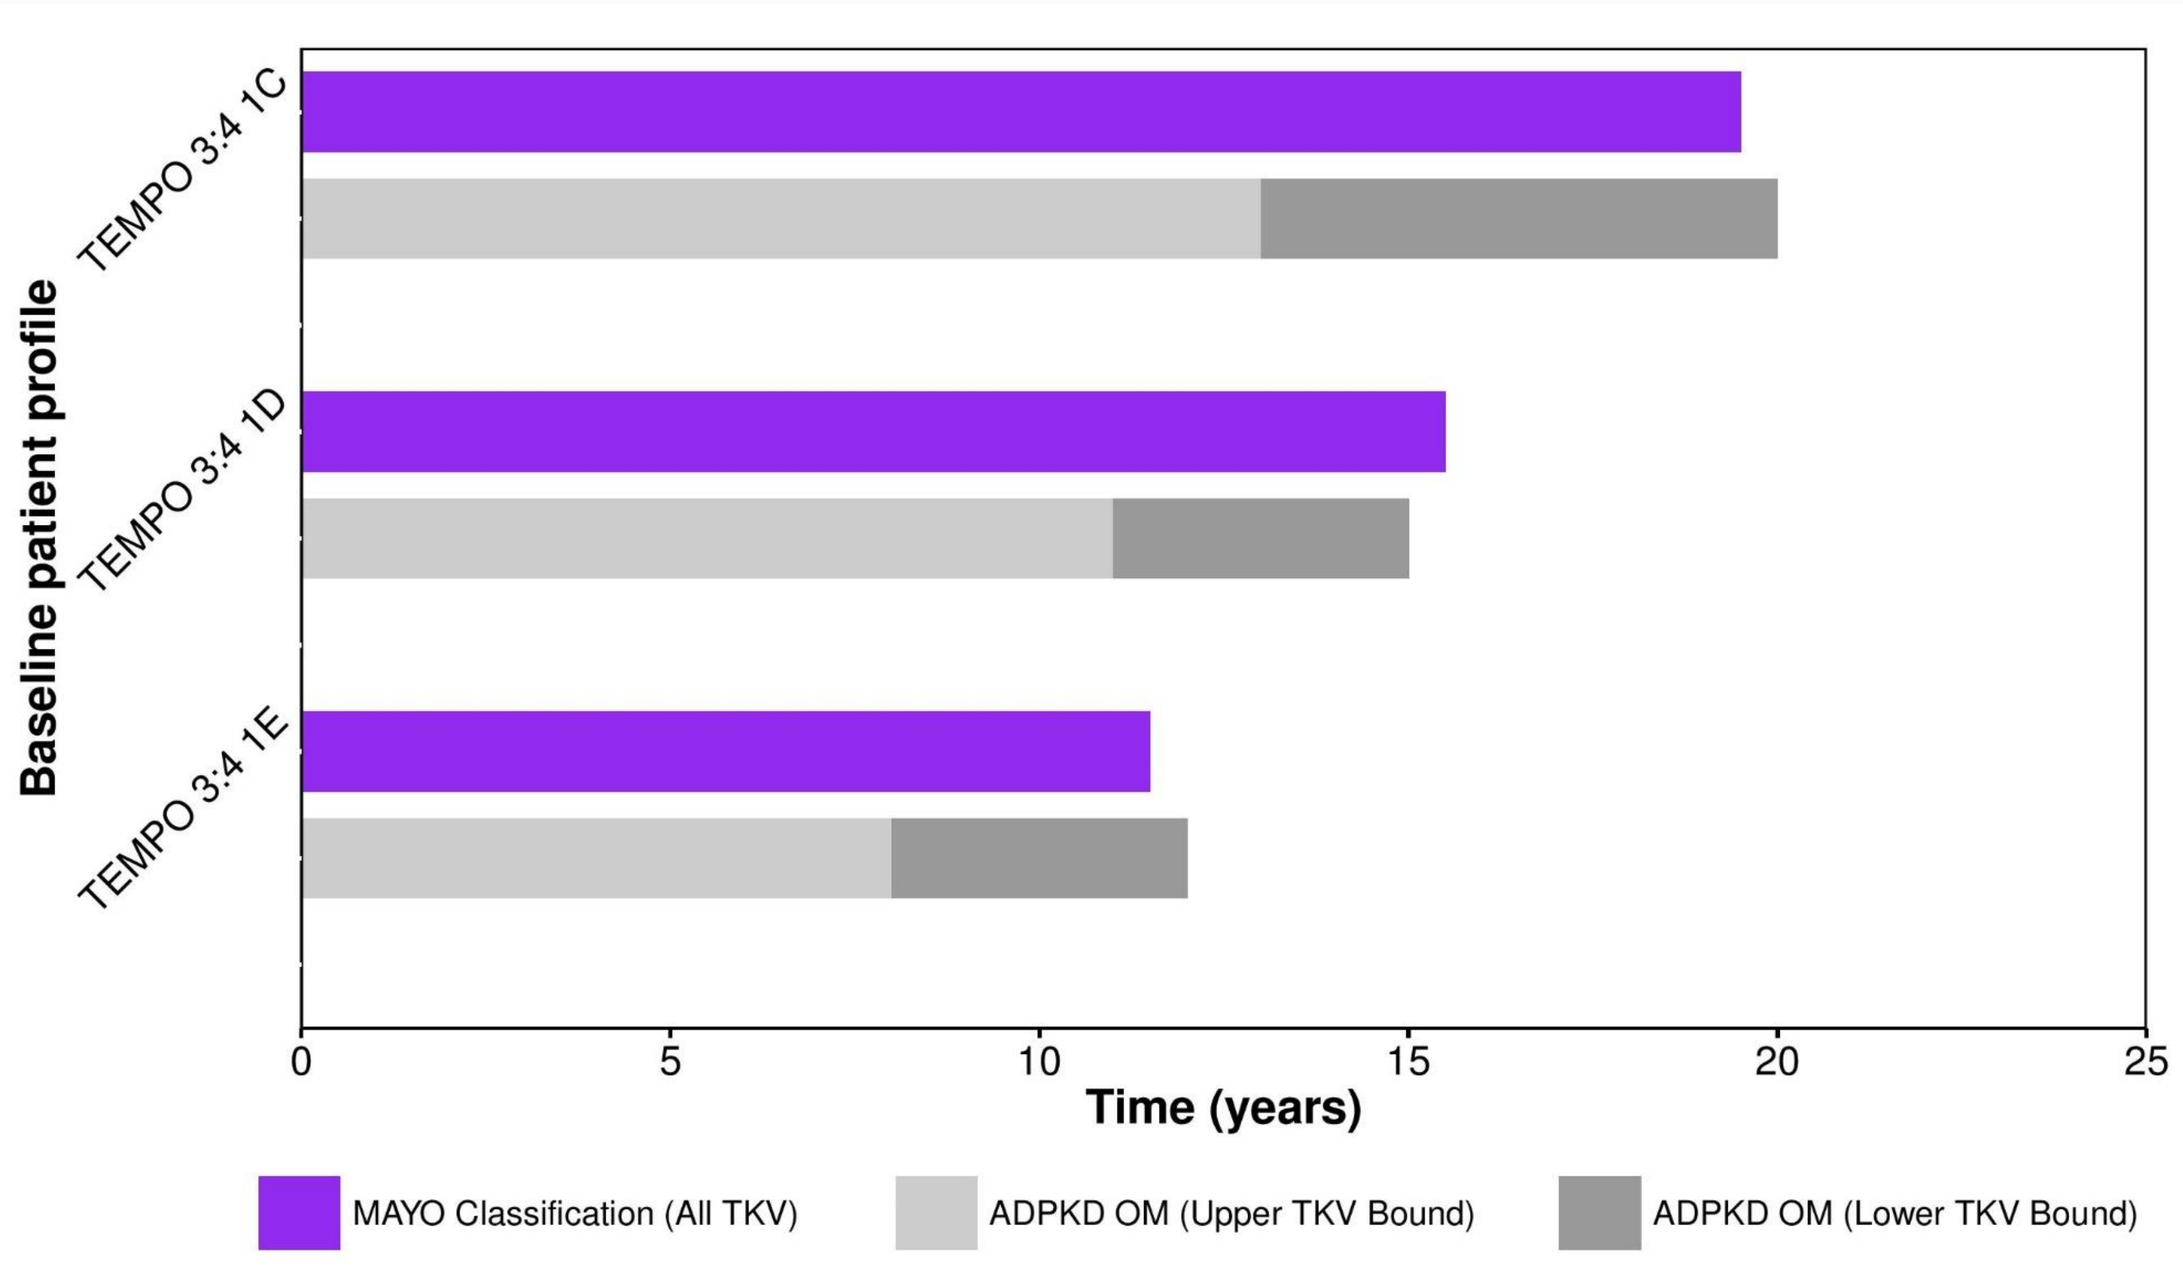

TEMPO 3:4 1C
TEMPO 3:4 1D
TEMPO 3:4 1E
Baseline patient profile
Time (years)
MAYO Classification (All TKV)
ADPKD OM (Upper TKV Bound)
ADPKD OM (Lower TKV Bound)

Figure 2. Within-class variability in time to ESRD for TEMPO 3:4 mean subgroup profiles, predicted using the Mayo Classification and ADPKD OM (for TKV range)

Upper bound for 1E presented at TKV 5,000 ml

When initiated with the mean TEMPO 3:4 subgroup profiles, the vast majority of simulated patients were predicted to reach ESRD within their lifetime; the expected duration of dialysis and the predicted number of transplants conducted increased with decreased time to ESRD (profiles 1C to 1E) (Table 3).

CONCLUSIONS

Predictions of renal decline obtained using the Mayo classification and ADPKD OM were consistent. Differences in the results of the two approaches may reflect heterogeneity of study population and modelling methods.

The ADPKD OM complements the methodology of the Mayo classification, enabling the identification of patients at highest risk for rapid renal decline.

The ability to model long-term ESRD burden and the impact of therapeutic intervention may deliver additional insight to aid planning of healthcare service delivery and the optimal timing of treatment initiation, thus maximising patient benefit.

REFERENCES

1. Spithoven EM, et al. Nephrol Dial Transplant. 2014 Sep;29 Suppl 4:iv15-25.
2. Schrier RW, et al. J Am Soc Nephrol 2014; 25: 2399-2418.
3. Woon C, et al. BMC Nephrol. 2015 Aug 15;16:140
4. Irazabal MV, et al. J Am Soc Nephrol. 2015 Jan;26(1):160-72
5. Gansevoort RT, et al. Nephrol Dial Transplant. 2016 Mar;31(3):337-48
6. Robinson P, et al. Development of a model to predict disease progression in autosomal dominant polycystic kidney disease (ADPKD). European Renal Association/European Dialysis and Transplant Association (ERA/EDTA) 2015.
7. Torres VE, et al. N Engl J Med 2012; 367: 2407-2418.
8. Robinson P, et al. Assessing the long-term outcomes of autosomal dominant polycystic kidney disease (ADPKD) using the ADPKD Outcomes Model: a UK case study. European Renal Association/European Dialysis and Transplant Association (ERA/EDTA) 2015.
9. Irazabal MV, et al. Prognostic Enrichment Design in Clinical Trials for ADPKD: The TEMPO 3:4 Clinical Trial. American Society of Nephrology (ASN) 2015.

Presented at 53rd ERA-EDTA Congress, Vienna, 21st-23rd May 2016

53 ERA 13--SP

Renal development and cystic diseases.

Karl OReilly

DOI: 10.3252/pso.eu.53era.2016

ePosters supported by F. Hoffmann- La Roche Ltd.