Vifor Fresenius Medical Care Renal Pharma

PHARMACOECONOMIC ASSESSMENT OF PATIENTS SWITCHING TO SUCROFERRIC OXHYDROXIDE FROM SEVELAMER CARBONATE IN PATIENTS ON HEMODIALYSIS IN SEVEN EUROPEAN COUNTRIES

Connolly M[1], Kotsopoulos N[1], Vrouchou P[2], Walpen S[3]

1.Global Market Access Solutions, Saint-Prex, Switzerland,
2.Vifor Pharma, Glattbrugg, Switzerland
3.Vifor Fresenius Medical Care Renal Pharma, Glattbrugg, Switzerland

Background

- The majority of patients with late-stage chronic kidney disease (CKD) who are on hemodialysis (HD) develop hyperphosphatemia and require oral phosphate binders to reduce the absorption of dietary phosphate.[1,2]
- Phosphate binders must be taken with every meal, often in conjunction with other medications or nutritional supplements, leading to a high pill burden (PB) for patients on dialysis.[3]
- Sucroferric oxyhydroxide (SFOH) is a non-calcium, iron-based phosphate binder indicated for the control of serum phosphorus (sPhos) levels in adult CKD patients on HD or peritoneal dialysis (PD).
- SFOH has demonstrated similar efficacy and tolerability to sevelamer carbonate (SEV), but with a lower PB, in Phase 3 studies.[4,5] Recent UK economic analyses of SFOH, alongside its clinical trials, showed that compared to SEV, SFOH appears to be cost-effective.[6,7]
- A more recent US retrospective database analysis, among in-center HD patients who switched from SEV to SFOH, showed, after three and six months: patients with in-range sPhos (3.5-5.5 mg/dl) increased in those who switched from SEV, while PB also decreased with SFOH suggesting lower cost per patient responding for SFOH.[8]

Research Aim

- The objective of this study was to assess the 3-monthly and 6-monthly cost per responder for SFOH versus SEV in Austria, France, Germany, Italy, Spain, United Kingdom (UK), and Switzerland, using a retrospective, real-life data analysis.

Methods & Data

- Response rates and PB were obtained from the retrospective real-life study which assessed changes in sPhos and PB in HD patients prescribed SFOH through a renal pharmacy service over three and six months as part of routine care.[8]
- Number needed to treat (NNT) to achieve in-range sPhos (3.5-5.5 mg/dl) were estimated based on the observed response rates (i.e., NNT = 1: proportion of patients with in-range sPhos) and subsequently, the 6-monthly costs per responder were calculated for SFOH and SEV.
- Drug acquisition costs per pill of SFOH and SEV were obtained from official list prices and were weighted for local market shares.
- Three and 6-monthly treatment costs were calculated based on the weighted drug acquisition costs per day and the observed, in the real-life study, daily PB.[8]
- Sensitivity analysis was conducted for the PB based on the minimum and maximum PB/day for SFOH and SEV (±30% and ±50% for SFOH and SEV, respectively).

Results

- The retrospective real-life study showed a 74% and 98% increase in patients with in-range sPhos after three and six months, respectively.
- After three months, patients switching from SEV, with in-range sPhos (3.5-5.5 mg/dl), increased from 14.1% to 24.5% (74%) whereas, after 6 months, patients with in-range sPhos (3.5-5.5 mg/dl), increased to 27.9% (98%).[8]
- Overall, mean PB (pills/day) decreased from 10.1 to 3.8 (62% fewer pills from baseline to 3 months) to 3.9 pills (61% fewer pills from 3 to 6 months).[8]
- Based on these estimates the calculated NNT at three months was 4.1 and 7.0 for SFOH and SEV, respectively.
- At 6 months the corresponding NNT was 3.6 and 7 for SFOH and SEV, respectively.
- In the base case analysis (Figure 1 & Figure 2) SFOH appears to attain in-range sPhos (3.5-5.5 mg/dl) at a lower three and 6-monthly cost per patient compared to SEV across the countries included in this analysis.
- Sensitivity analysis (Figure 3) for PB suggests that SFOH is consistently associated with lower cost per responder.

Figure 1: 3-monthly cost per responder for SFOH and SEV

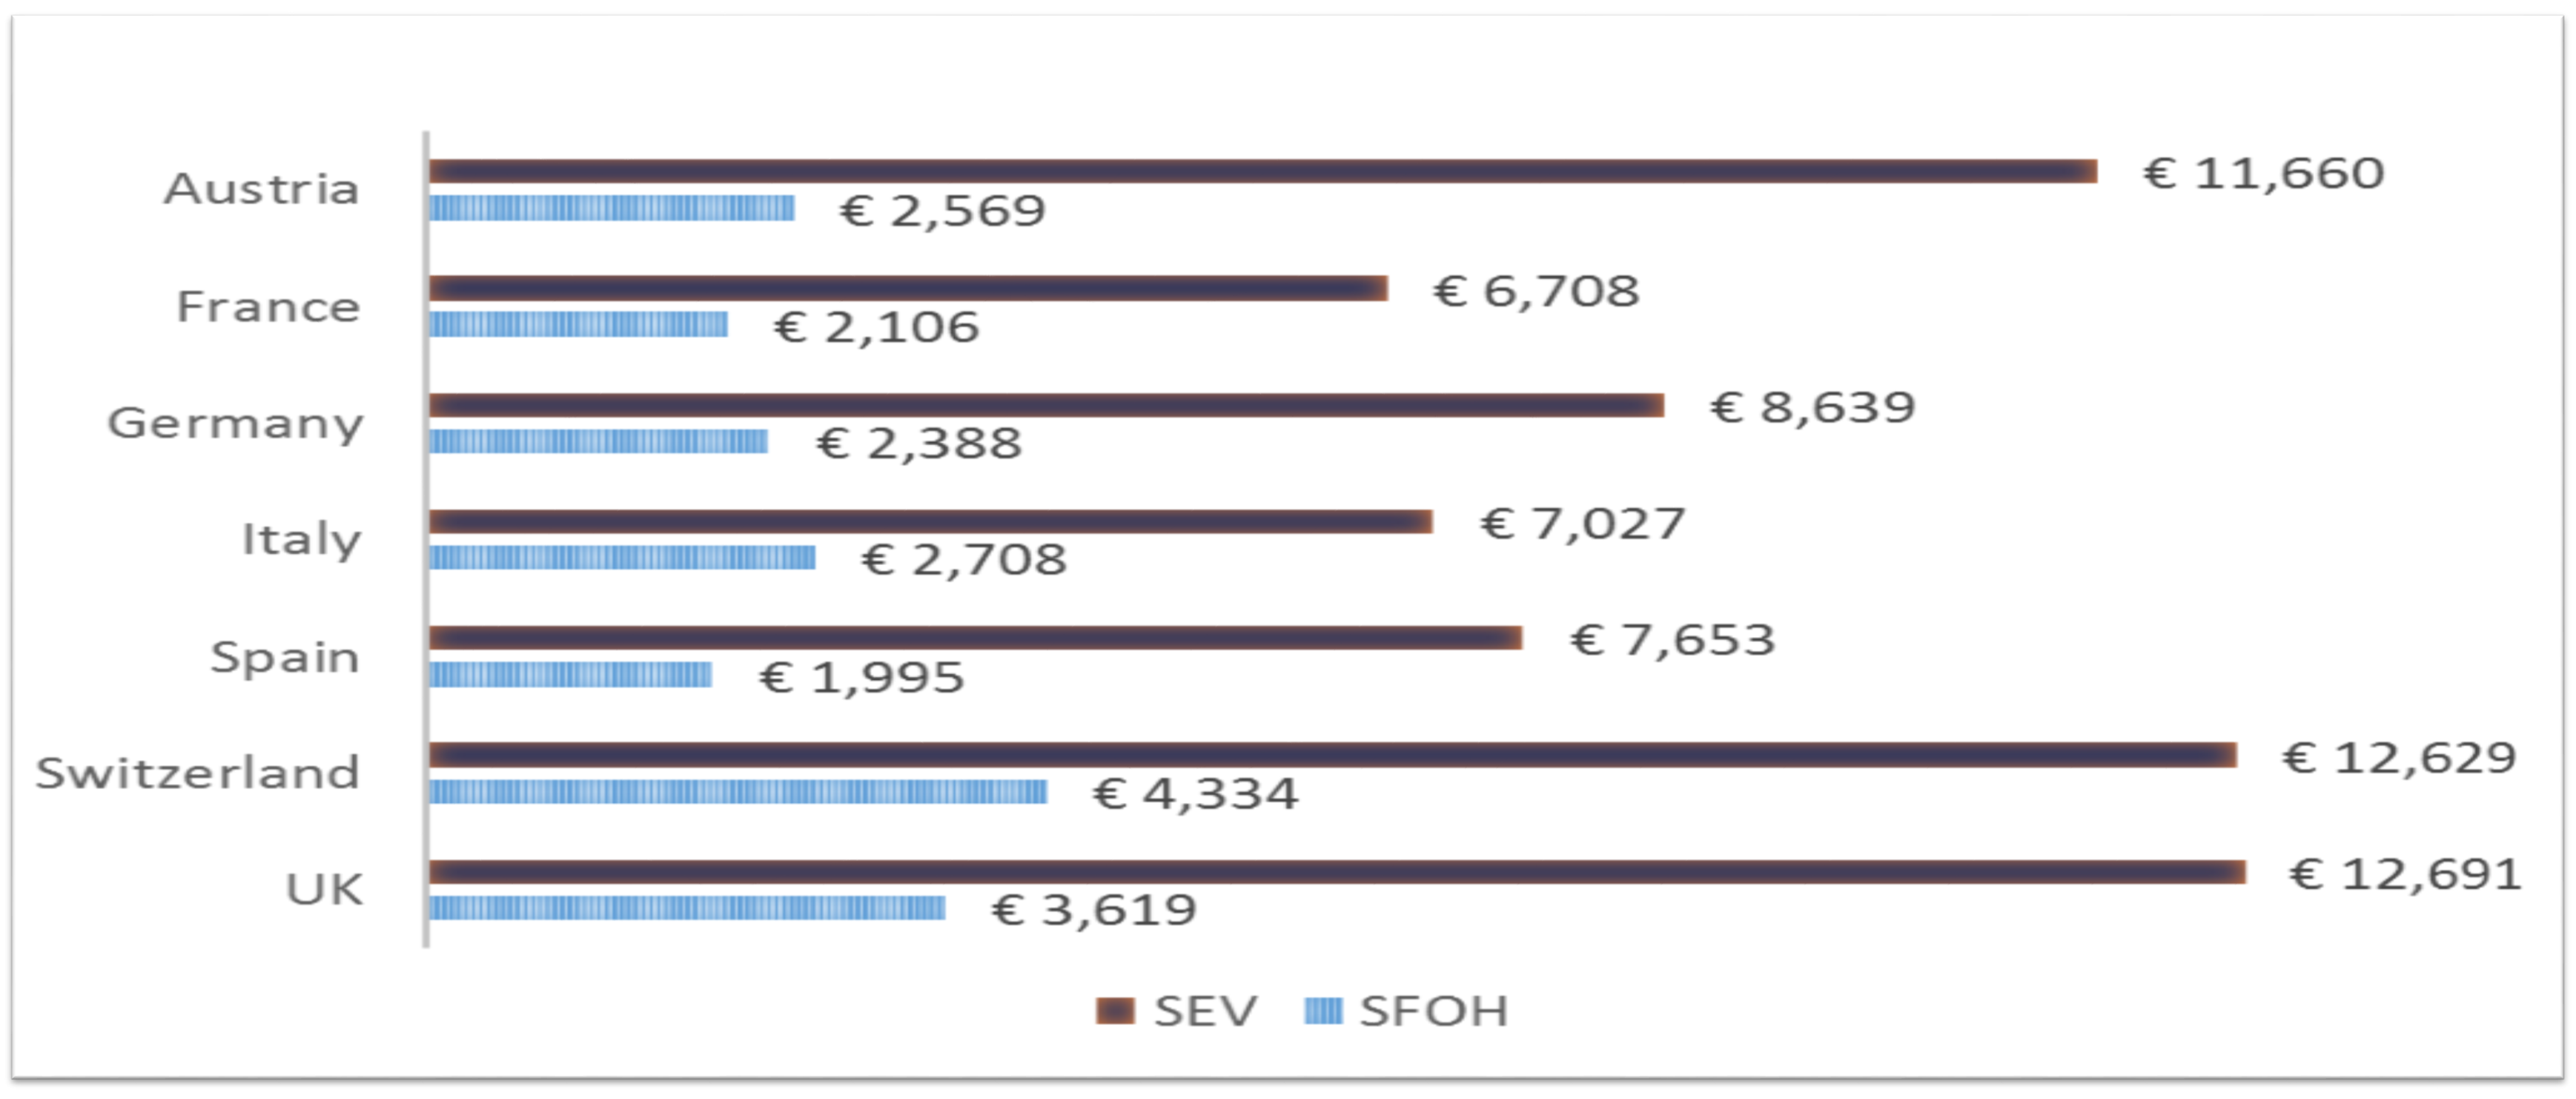
Austria
€ 11,660
€ 2,569
France
€ 6,708
€ 2,106
Germany
€ 8,639
€ 2,388
Italy
€ 7,027
€ 2,708
Spain
€ 7,653
€ 1,995
Switzerland
€ 12,629
€ 4,334
UK
€ 12,691
€ 3,619
SEV
SFOH

- At 3 months, SFOH was consistently less costly compared to SEV. The cost per responder difference between SFOH and SEV was the greatest in Austria with a difference of -9,091€, followed by UK at -9,072€, Switzerland at -8,295€, Germany at -6,252€, Spain at -5,659€, France at -4,502€, and Italy at -4,319€ .

Figure 2: 6-monthly cost per responder for SFOH and SEV

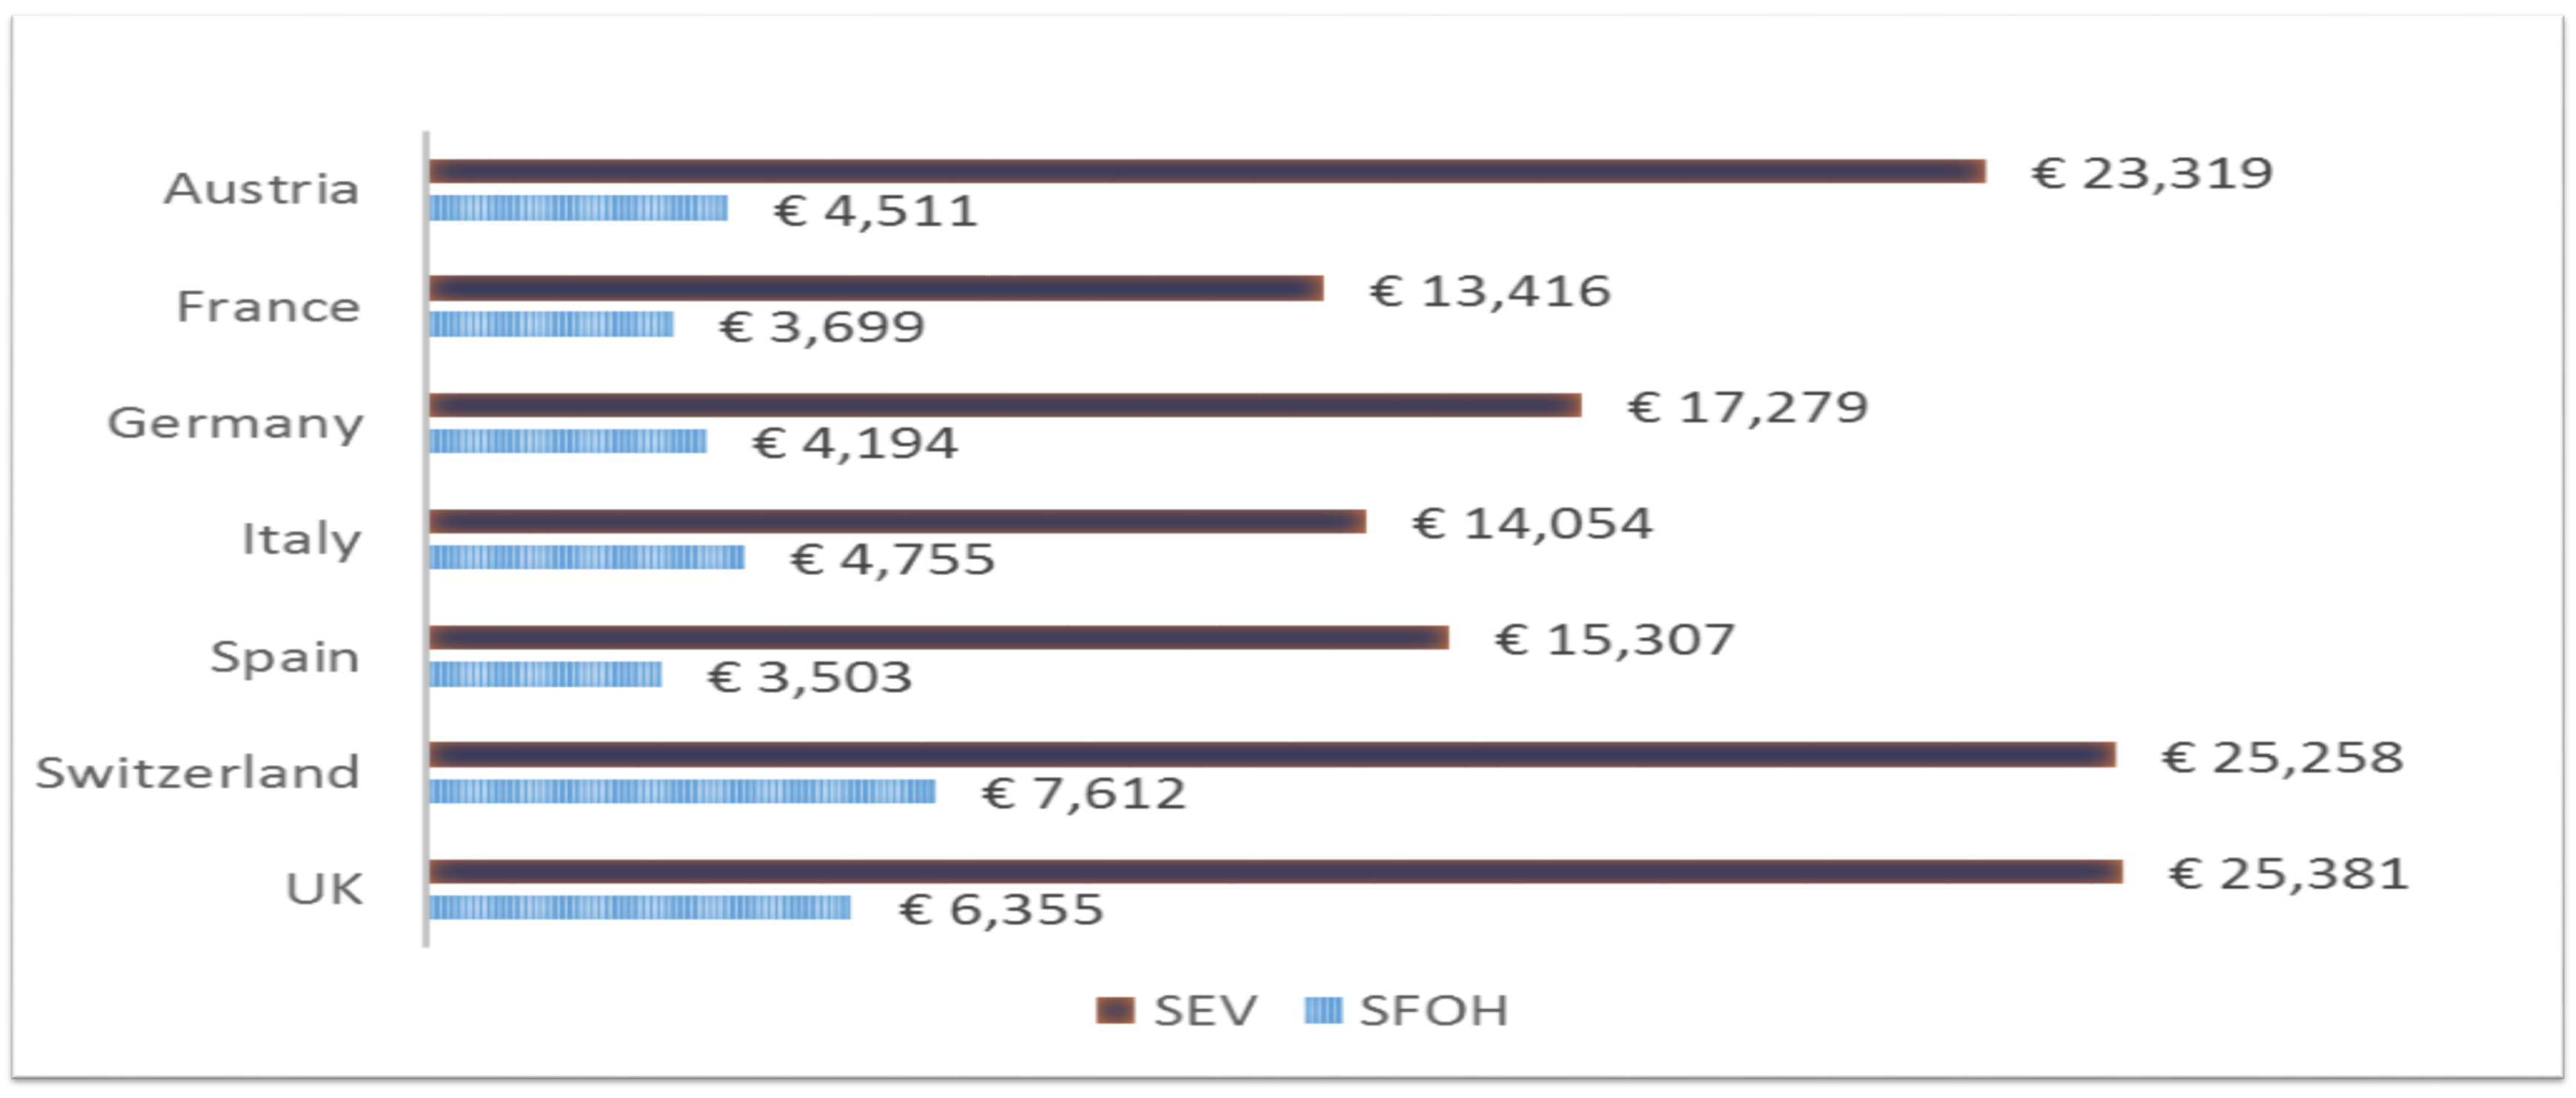
Austria
€ 23,319
€ 4,511
France
€ 13,416
€ 3,699
Germany
€ 17,279
€ 4,194
Italy
€ 14,054
€ 4,755
Spain
€ 15,307
€ 3,503
Switzerland
€ 25,258
€ 7,612
UK
€ 25,381
€ 6,355
SEV
SFOH

- At 6 months, SFOH was consistently less costly compared to SEV. The cost per responder difference between SFOH and SEV was the greatest in UK with a difference of -19,026€, followed by Austria at -18,808€, Switzerland at -17,647€, Germany at -13,085€, Spain at -11,804€, France at -9,717€, and Italy at -9,299€.

Figure 3: Sensitivity analysis: Effect of variating PB/day on differential cost per responder (SFOH vs. SEV)

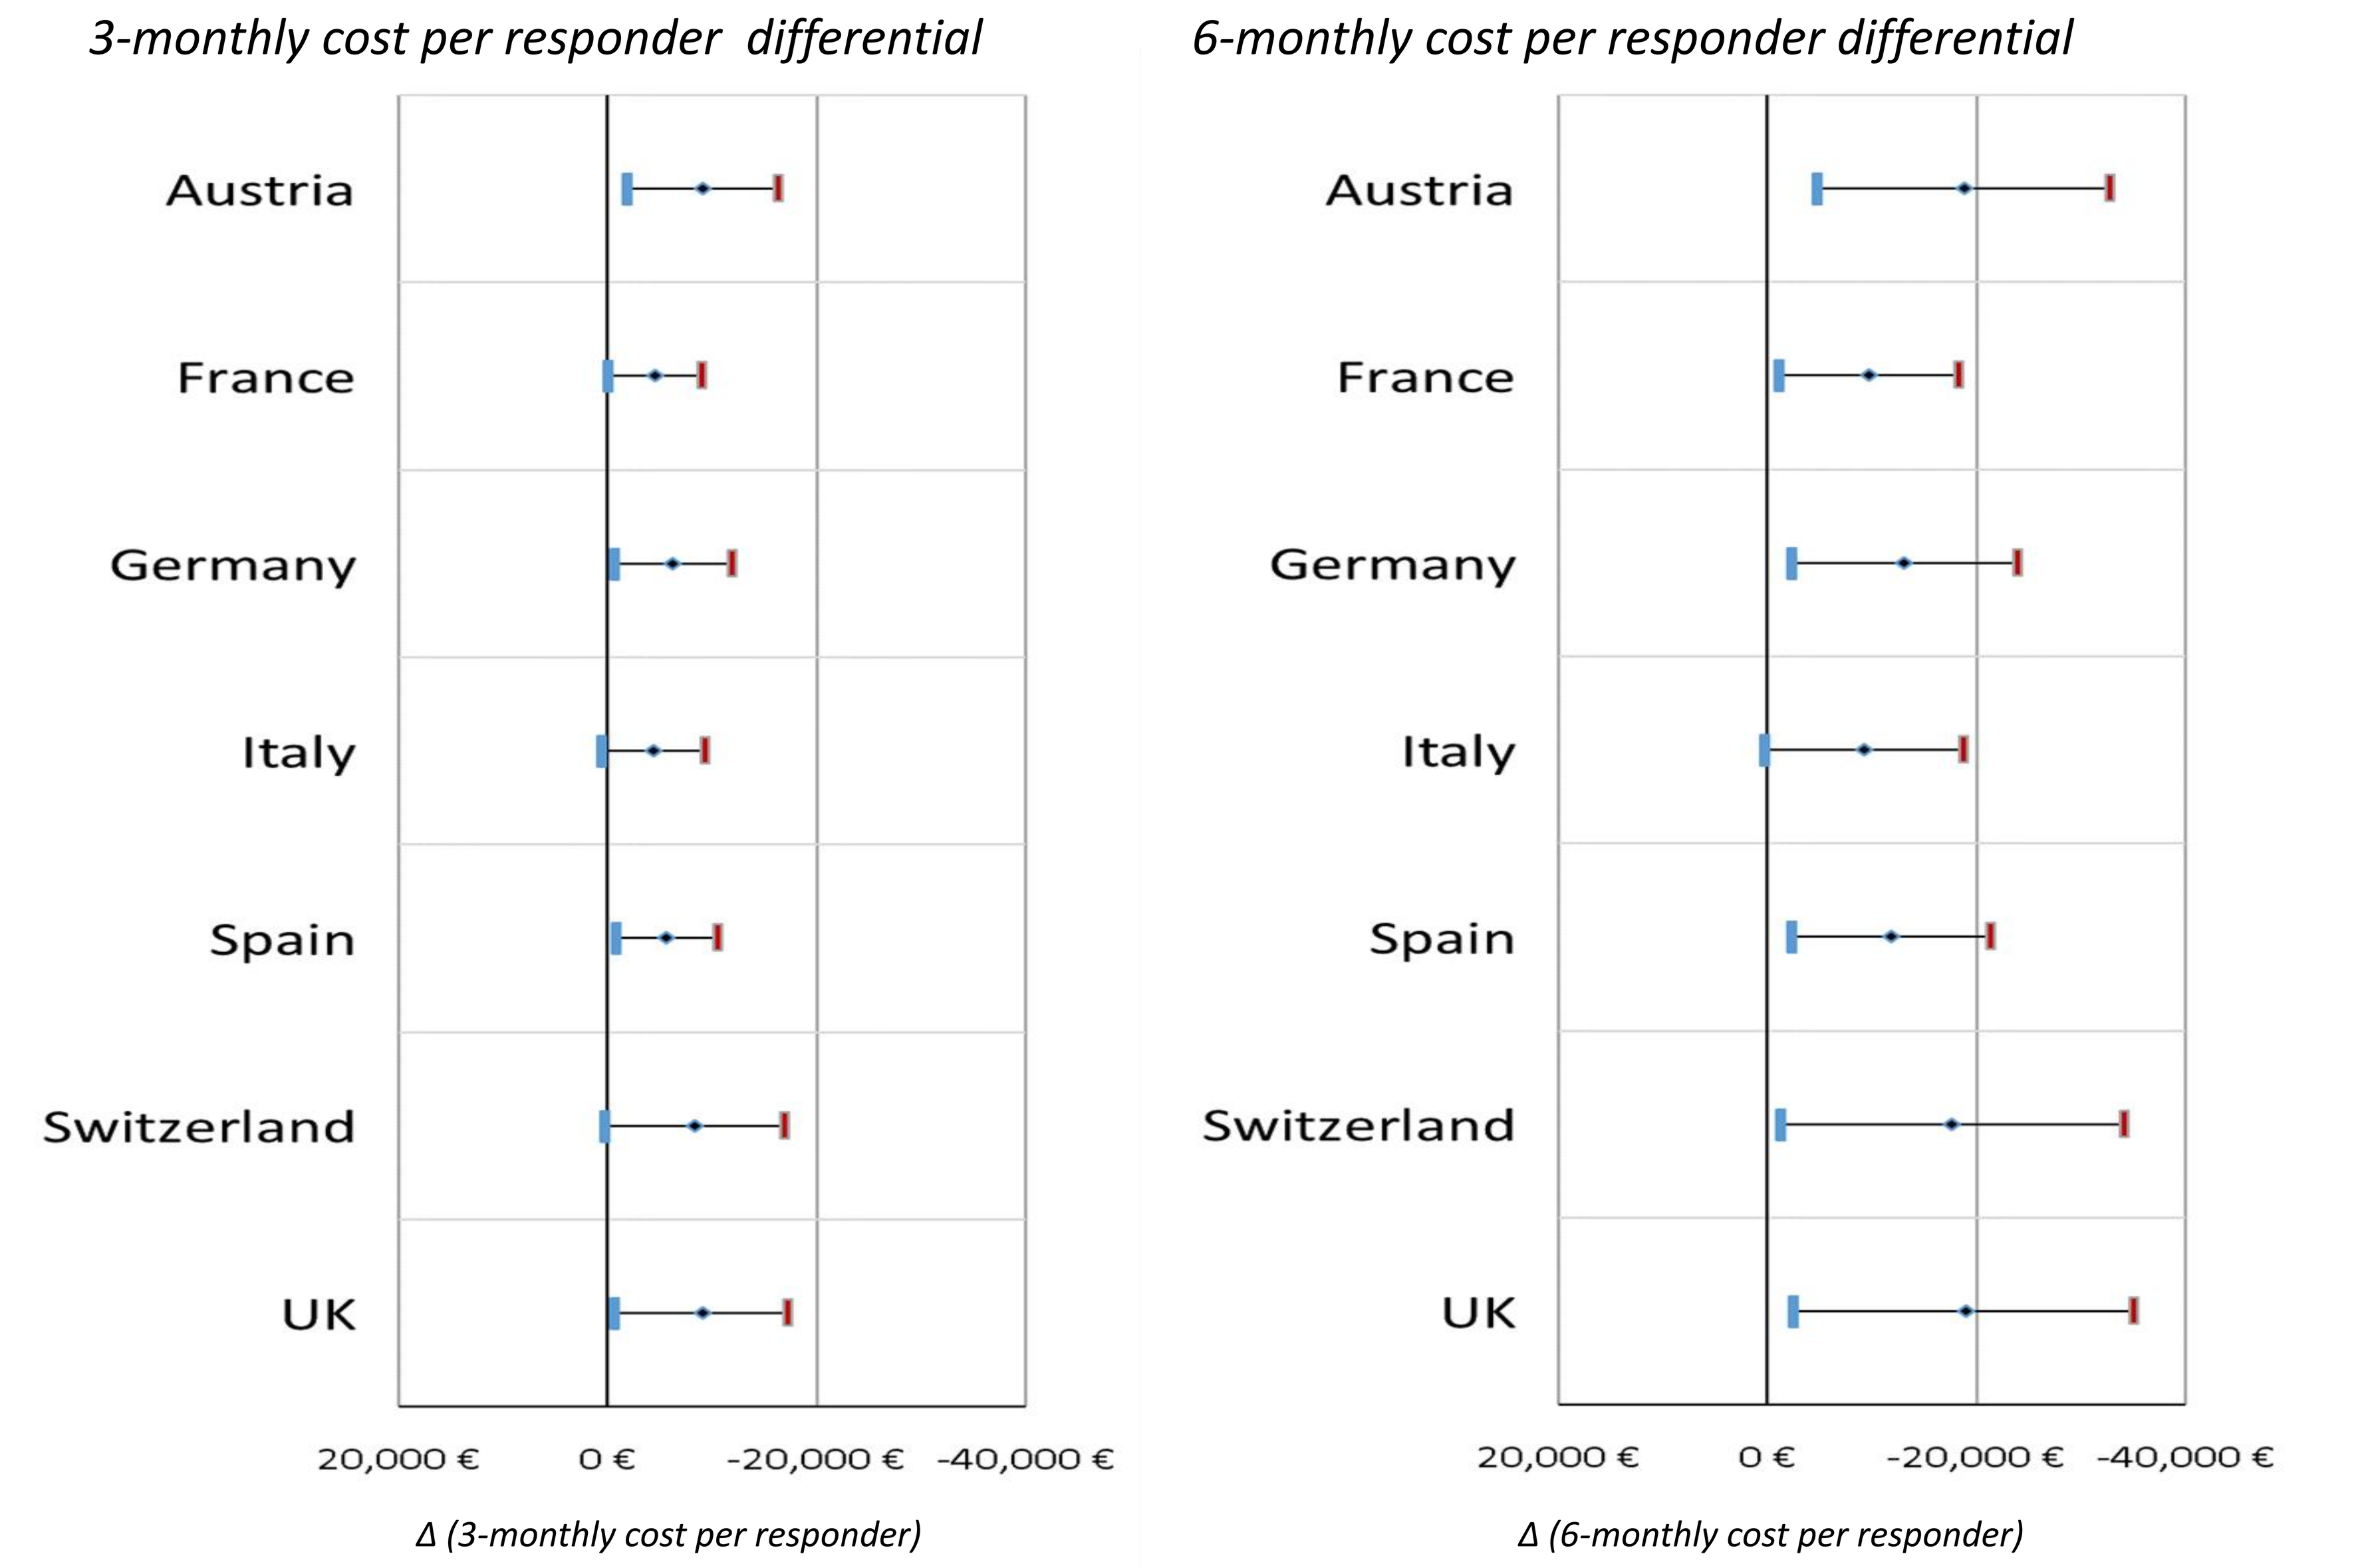
3-monthly cost per responder differential
6-monthly cost per responder differential
Austria
France
Germany
Italy
Spain
Switzerland
UK
20,000 €
0 €
-20,000 €
-40,000 €
Δ (3-monthly cost per responder)
Δ (6-monthly cost per responder)

- The 3 and 6-monthly costs per responder remained favorable for SFOH when the PB/day of the products under study were varied. Worst case scenario: SFOH PB +50% & SEV PB -50%. Base case scenario: SFOH 3.9 pills/day & SEV 10.1 pills/day. Best case scenario: SFOH PB -50% & SEV PB +50%.

References

1. Hutchison AJ. Kidney international. 2009; 75:906-914; 2. Locatelli F et al., Expert opinion on drug safety. 2014; 13:551-561; 3. Chiu YW et al., Clinical journal of the American Society of Nephrology: CJASN. 2009; 4:1089-1096; 4. Floege J, et al. Kidney Int. 2014;86:638-47; 5. Sprague S, et al. 2013; Presented at: American Society of Nephrology Kidney; 6. Gutzwiller FS et al., Pharmacoeconomics. 2015 Dec;33(12):1311-24. doi: 10.1007/s40273-015-0320-9; 7. SMC No. (1035/15) 8. Ficociello L. et al. NKF 2016

Conclusions

- SFOH appears to have a favorable NNT, and to attain the clinical target of in-range sPhos at a lower cost compared to SEV, suggesting favorable cost-effectiveness.
- This finding was consistently observed across all seven countries assessed at three and six months.
- The cost per responder remained favorable for SFOH when the relative PB (and hence costs) of the products under study were varied.

54 ERA
753--MP
Dialysis - Health service
Mark Connolly
DOI: 10.3252/pso.eu.54ERA.2017

ePosters supported by F. Hoffmann- La Roche Ltd.
PosterSessionOnline.com