WFH 2024 WORLD CONGRESS
APRIL 21-24 MADRID, SPAIN

athn transcends

ATHN Transcends: Hemophilia Gene Therapy Outcomes Arm – A Long-Term Follow-Up Study on the Safety and Effectiveness of Gene Therapy in People with Hemophilia

Authors:

Janice M. Staber, MD,[1] Blanca Salazar, MD,[2] Roberto Guillen-Gonzalez, MD,[2] Michael Recht, MD, PhD, MBA,[3,4] Tammuella Chrisentery-Singleton, MD,[5,6] Ulrike M. Reiss, MD[7]

[1]Iowa Hemophilia and Thrombosis Center, University of Iowa Stead Family Children's Hospital; [2]CSL Behring; [3]National Blood Disorders Foundation; [4]Yale University School of Medicine; [5]American Thrombosis and Hemostasis Network; [6]Ochsner Clinic Foundation; [7]Hemophilia Treatment Center, St. Jude's Children's Research Hospital

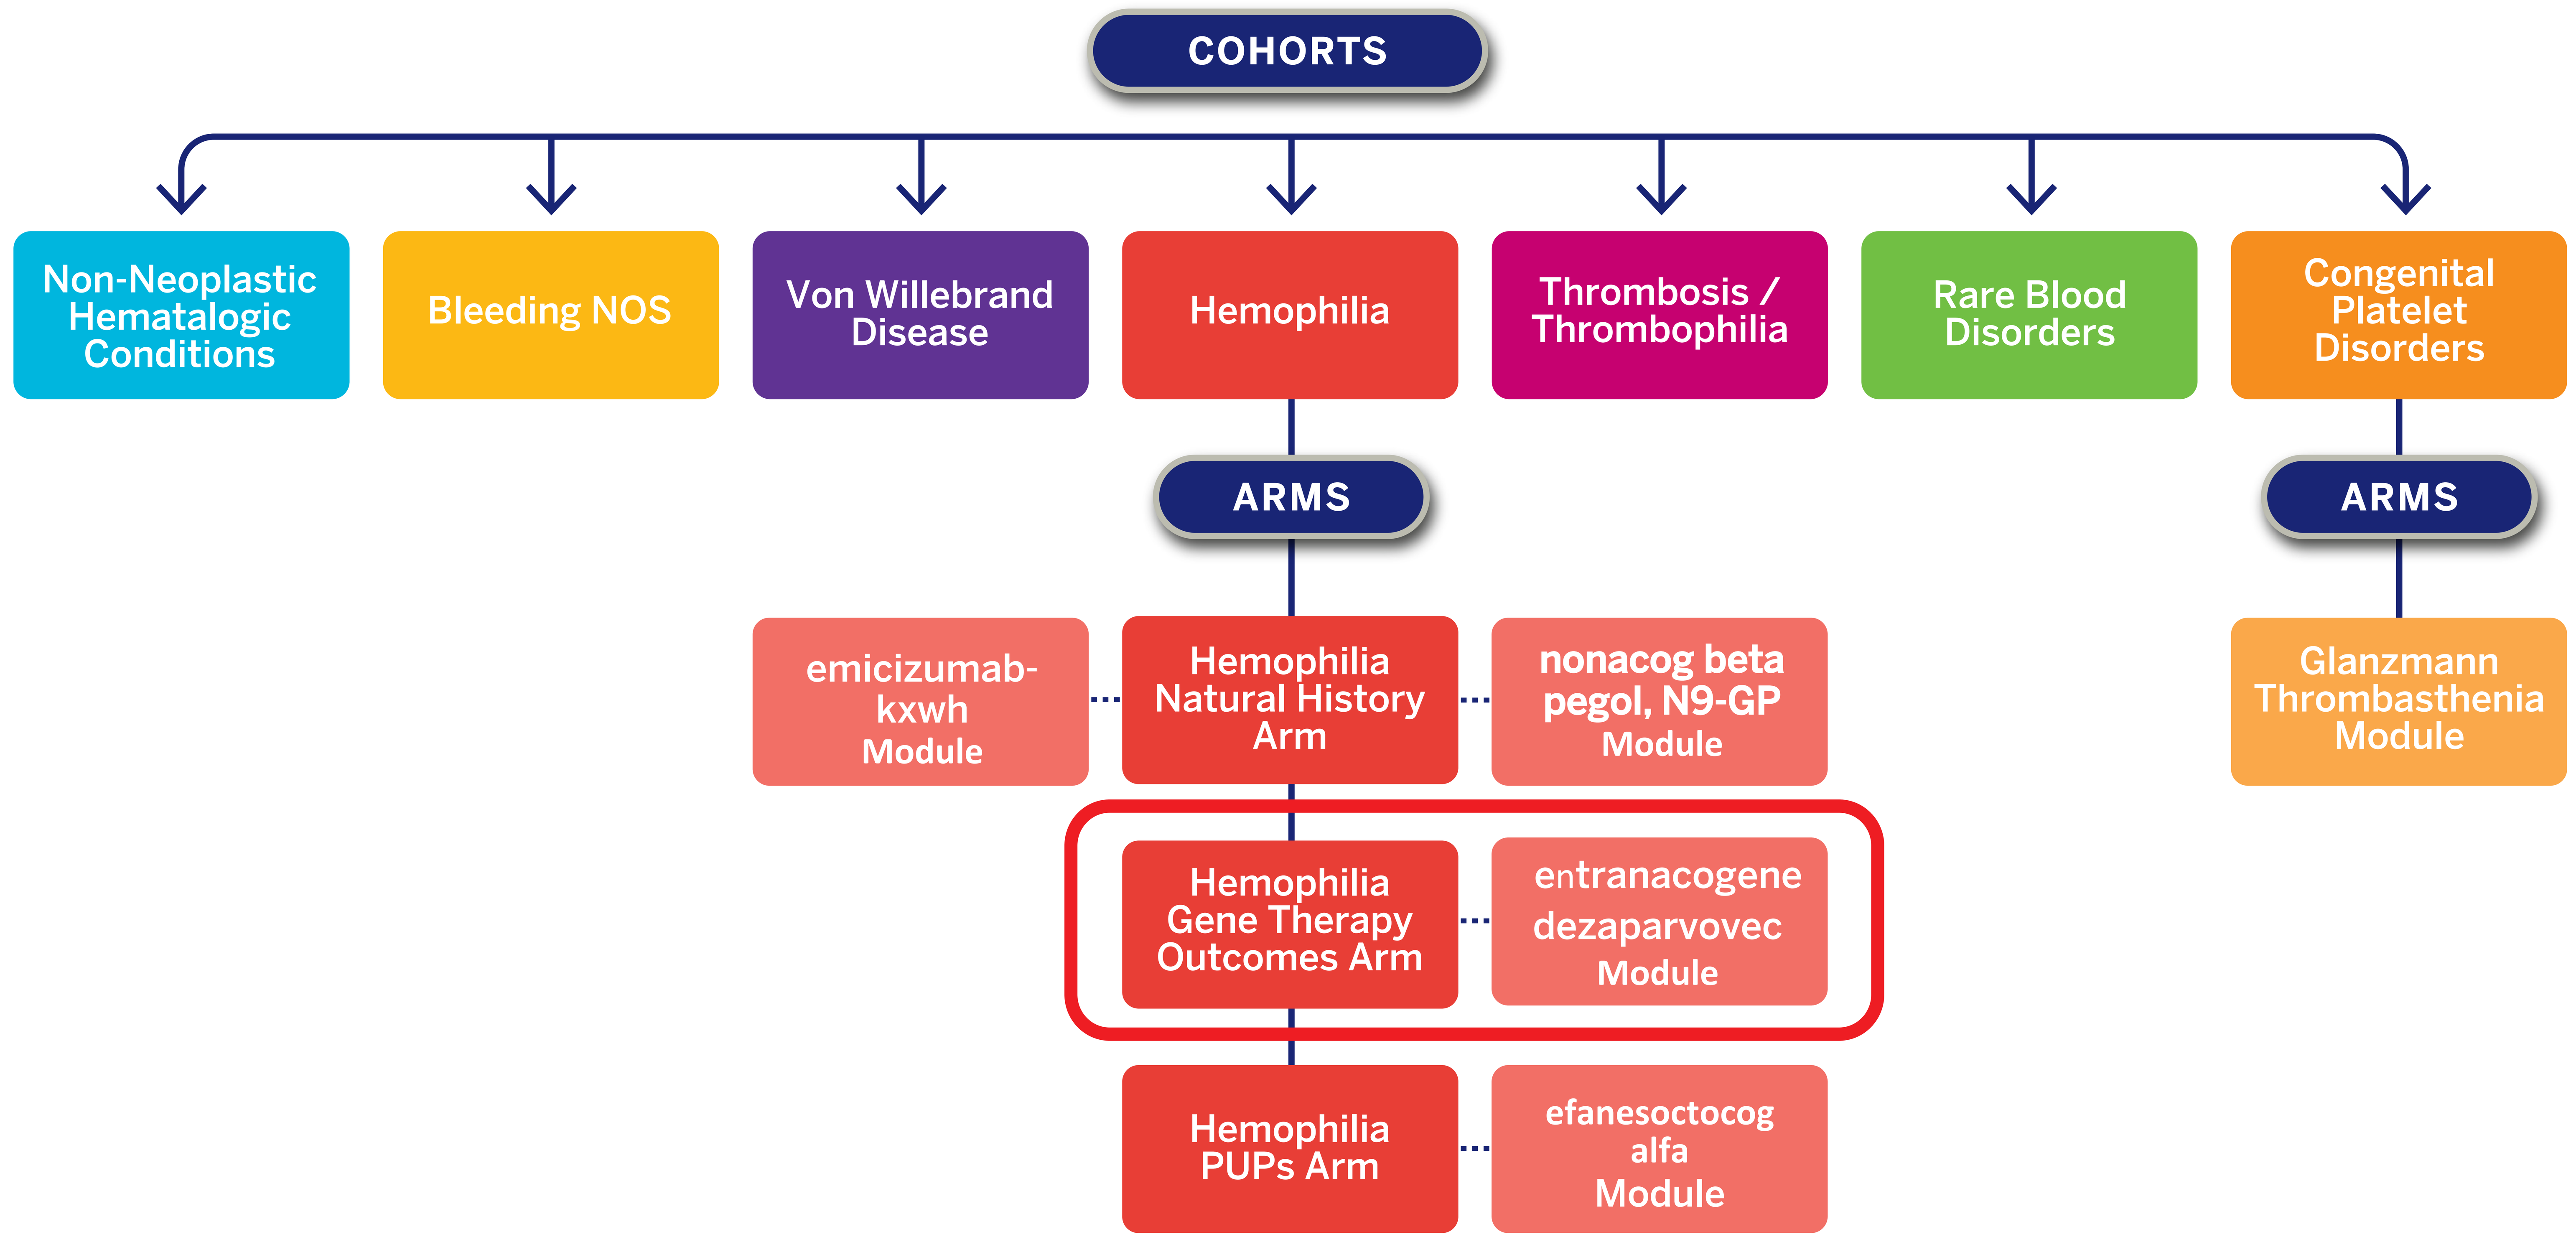

COHORTS
Non-Neoplastic Hematologic Conditions
Bleeding NOS
Von Willebrand Disease
Hemophilia
Thrombosis / Thrombophilia
Rare Blood Disorders
Congenital Platelet Disorders
ARMS
ARMS
emicizumab-kxwh Module
Hemophilia Natural History Arm
nonacog beta pegol, N9-GP Module
Glanzmann Thrombasthenia Module
Hemophilia Gene Therapy Outcomes Arm
entranacogene dezaparvovec Module
Hemophilia PUPs Arm
efanesoctocog alfa Module

Rationale

The Hemophilia Gene Therapy Outcomes Arm of ATHN Transcends assesses the safety and effectiveness of gene therapy. ATHN Transcends primary objective is to collect specific adverse events harmonized with the European Haemophilia Safety Surveillance (EUHASS). A module in the Hemophilia Gene Therapy Outcomes Arm, the entranacogene dezapar-vovec Module, is an observational, post-authorization, long-term, follow up, multicenter study to investigate the short-and long-term ef-fectiveness and safety of etranocogene dezaparvovec treatment in adults with hemophilia B in the United States.

Objectives

Hemophilia Gene Therapy Outcomes Arm Objectives:

To determine the safety and effectiveness of adeno-associated viral vector (AAV) or lentiviral vector (LV)-mediated factor VIII (FVIII) and factor IX (FIX) therapies used for participants with hemophilia for at least 15 years after vector infusion.

HEMGENIX® Module Objectives:

To investigate the short- and long-term safety and effectiveness profile of entracogene dezaparvovec by following adults with hemophilia B who are treated with entracogene dezaparvovec or are on continuous FIX prophylax-is for 15 years.

Study Design

In the Hemophilia Gene Therapy Outcomes Arm, data will be collected as follows: once at time of enrollment and then every 3 months during the first year; then every 6 months and annually thereafter until year 15 after vector infusion. The total study duration is expected to be 20 years.

The entracogene dezaparvovec Module will collect data from 2 patient cohorts for analysis:

1. People with hemophilia B treated with etranocogene dezaparvovec in the United States.
2. People with hemophilia B on FIX prophylaxis and enrolled in the ATHN Transcends study.

Data Analysis

Most of the study arm outcome variables are discrete in nature, such as mortality, newly developed inhibitor, bleeding rate, etc. Some outcome measurements will be treated as continuous, like health-related quality of life. For each categorical variable, the frequency and percentage will be reported. In terms of a continuous measurement, the mean, median, standard deviation, interquartile range, minimum, and maximum values will be disclosed. During the study arm, the steering committee will evaluate the appropriateness of various statistical approaches based on the amount and quality of data collected. Data from participants who are enrolled in the arm, but never receive treatment with a gene therapy product, will only be included in analysis of relevant arm objectives.

Eligibility

Eligibility criteria: Includes participants from clinical trials, people considering gene therapy, and people treated with approved products. There are no exclusion criteria.

Summary

The ATHN Transcends: Hemophilia Gene Therapy Outcomes Arm is the only comprehensive follow-up registry study in the US for people with hemophilia B considering or after gene therapy:

- Long-term follow-up after gene therapy is critical to confirm short-term findings from trials and identify the unknowns.
- This study arm includes post-marketing required evaluations by data collection and analysis via product-specific modules (such as for entracogene dezaparvovec) and other future FDA-approved gene therapy products.
- ATHN Transcends has received central IRB approval and is currently being rolled out across participating ATHN Affiliates in the United States. Enrollment in the Hemophilia Gene Therapy Outcomes Arm and the etranacogene dezaparvovec Module can begin as soon as a site opens ATHN Transcends to enrollment.

To establish this study at your site, contact **support@athn.org**.

Conclusion

The ATHN Transcends: Hemophilia Gene Therapy Outcomes Arm provides a real-world mechanism to determine the safety of adeno-associated viral vector (AAV)-mediated factor FVIII and FIX therapies when used for participants with hemophilia B.

The Hemophilia Gene Therapy Outcomes Arm: etranacogene dezaparvovec Module of ATHN Transcends will contribute to updating the effectiveness and safety profile of etranacogene dezaparvovec in a larger population of patients through the collection of Good Clinical Practice-grade data.

The ATHN Transcends Hemophilia Gene Therapy Outcomes Arm and the entranacogene dezaparrvovec **Module are open for enrollment.**

References

Pipe SW, Leebeek FWG, Recht M, Key NS, et al. Gene Therapy with Etranacogene Dezaparvovec for Hemophilia B. N Engl J Med 2023;388:706-18. DOI: 10.1056/NEJMoa2211644

Drygalski A, Gomez E, Giermasz A, Castaman G, et al. Stable and durable factor IX levels in hemophilia B patients over 3 years post etranacogene dezaparvovec gene therapy. Blood Advances. 2022. https://pubmed.ncbi.nlm.nih.gov/36490302/

Poster # PO-025