WFH 2024 WORLD CONGRESS
APRIL 21-24 MADRID, SPAIN

athn transcends

ATHN Transcends: Natural History Cohort Study of Bleeding Symptoms and Treatment Outcomes in Patients with Glanzmann Thrombasthenia

Authors:
Divyaswathi Citla Sridhar[1], Sanjay Ahuja[2], Tammuella Chrisentery-Singleton[3,4], Carrie O'Neill[3], Catherine Rea[5], Jigar Amin[5], Michael Recht[6,7], Meera Chitlur[8]
[1]Arkansas Children's Hospital; [2]Rainbow Babies & Children's Hospital, Case Western Reserve University; [3]American Thrombosis and Hemostasis Network; [4]Ochsner Clinic Foundation; [5]Hemab Therapeutics; [6]National Bleeding Disorders Foundation; [7]Yale University School of Medicine; [8]Central Michigan University, Children's Hospital of Michigan

Background

The ATHN Transcends Congenital Platelet Disorders (CPD) Natural History Arm is investigating the natural history of the safety and effectiveness of hemostatic therapies in the prevention or treatment of bleeding events in adults and children with congenital platelet disorders. It is a longitudinal, prospective, retrospective, observational cohort arm of ATHN Transcends. The Glanzmann Thrombasthenia (GT) Module is a registry in the Congenital Platelet Disorders Natural History Arm of ATHN Transcends, specific to patients with GT.

GT is a rare, inherited, qualitative platelet disorder due to deficiency of one or both platelet surface proteins alpha IIb beta 3. Reports of the impact of living with GT are lacking. Platelet transfusions which have been the mainstay of treatment for severe bleeding in people with GT, carry a number of risks including allergic reactions (anaphylaxis, acute lung injury), pathogen transmission, and the formation of allo-antibodies. As potential new therapies emerge, this registry allows unbiased, long term data collection on the safety and effectiveness of both current and new therapies in addition to information on the bleeding rate and phenotype, as well as describing their lived experience.

Rationale

Due to the rare nature of GT, data within individual center registries are limited. There is variation in the data held and method of collection. The aim of the study is to collect biospecimens, phenotypic characteristics, bleeding patterns, and treatment utilization, from people who have been diagnosed with GT. To achieve this, a comprehensive, multi-institutional, observational cohort study will be conducted to:

- Allow collection of longitudinal safety and clinical practice data for treatments in participants with GT, allowing monitoring of recently approved and well-established therapies;
- Aid new discovery, through the collection of biospecimens and phenotype data;
- Provide sufficient, unified data capture in participants with this rare disorder across the United States.

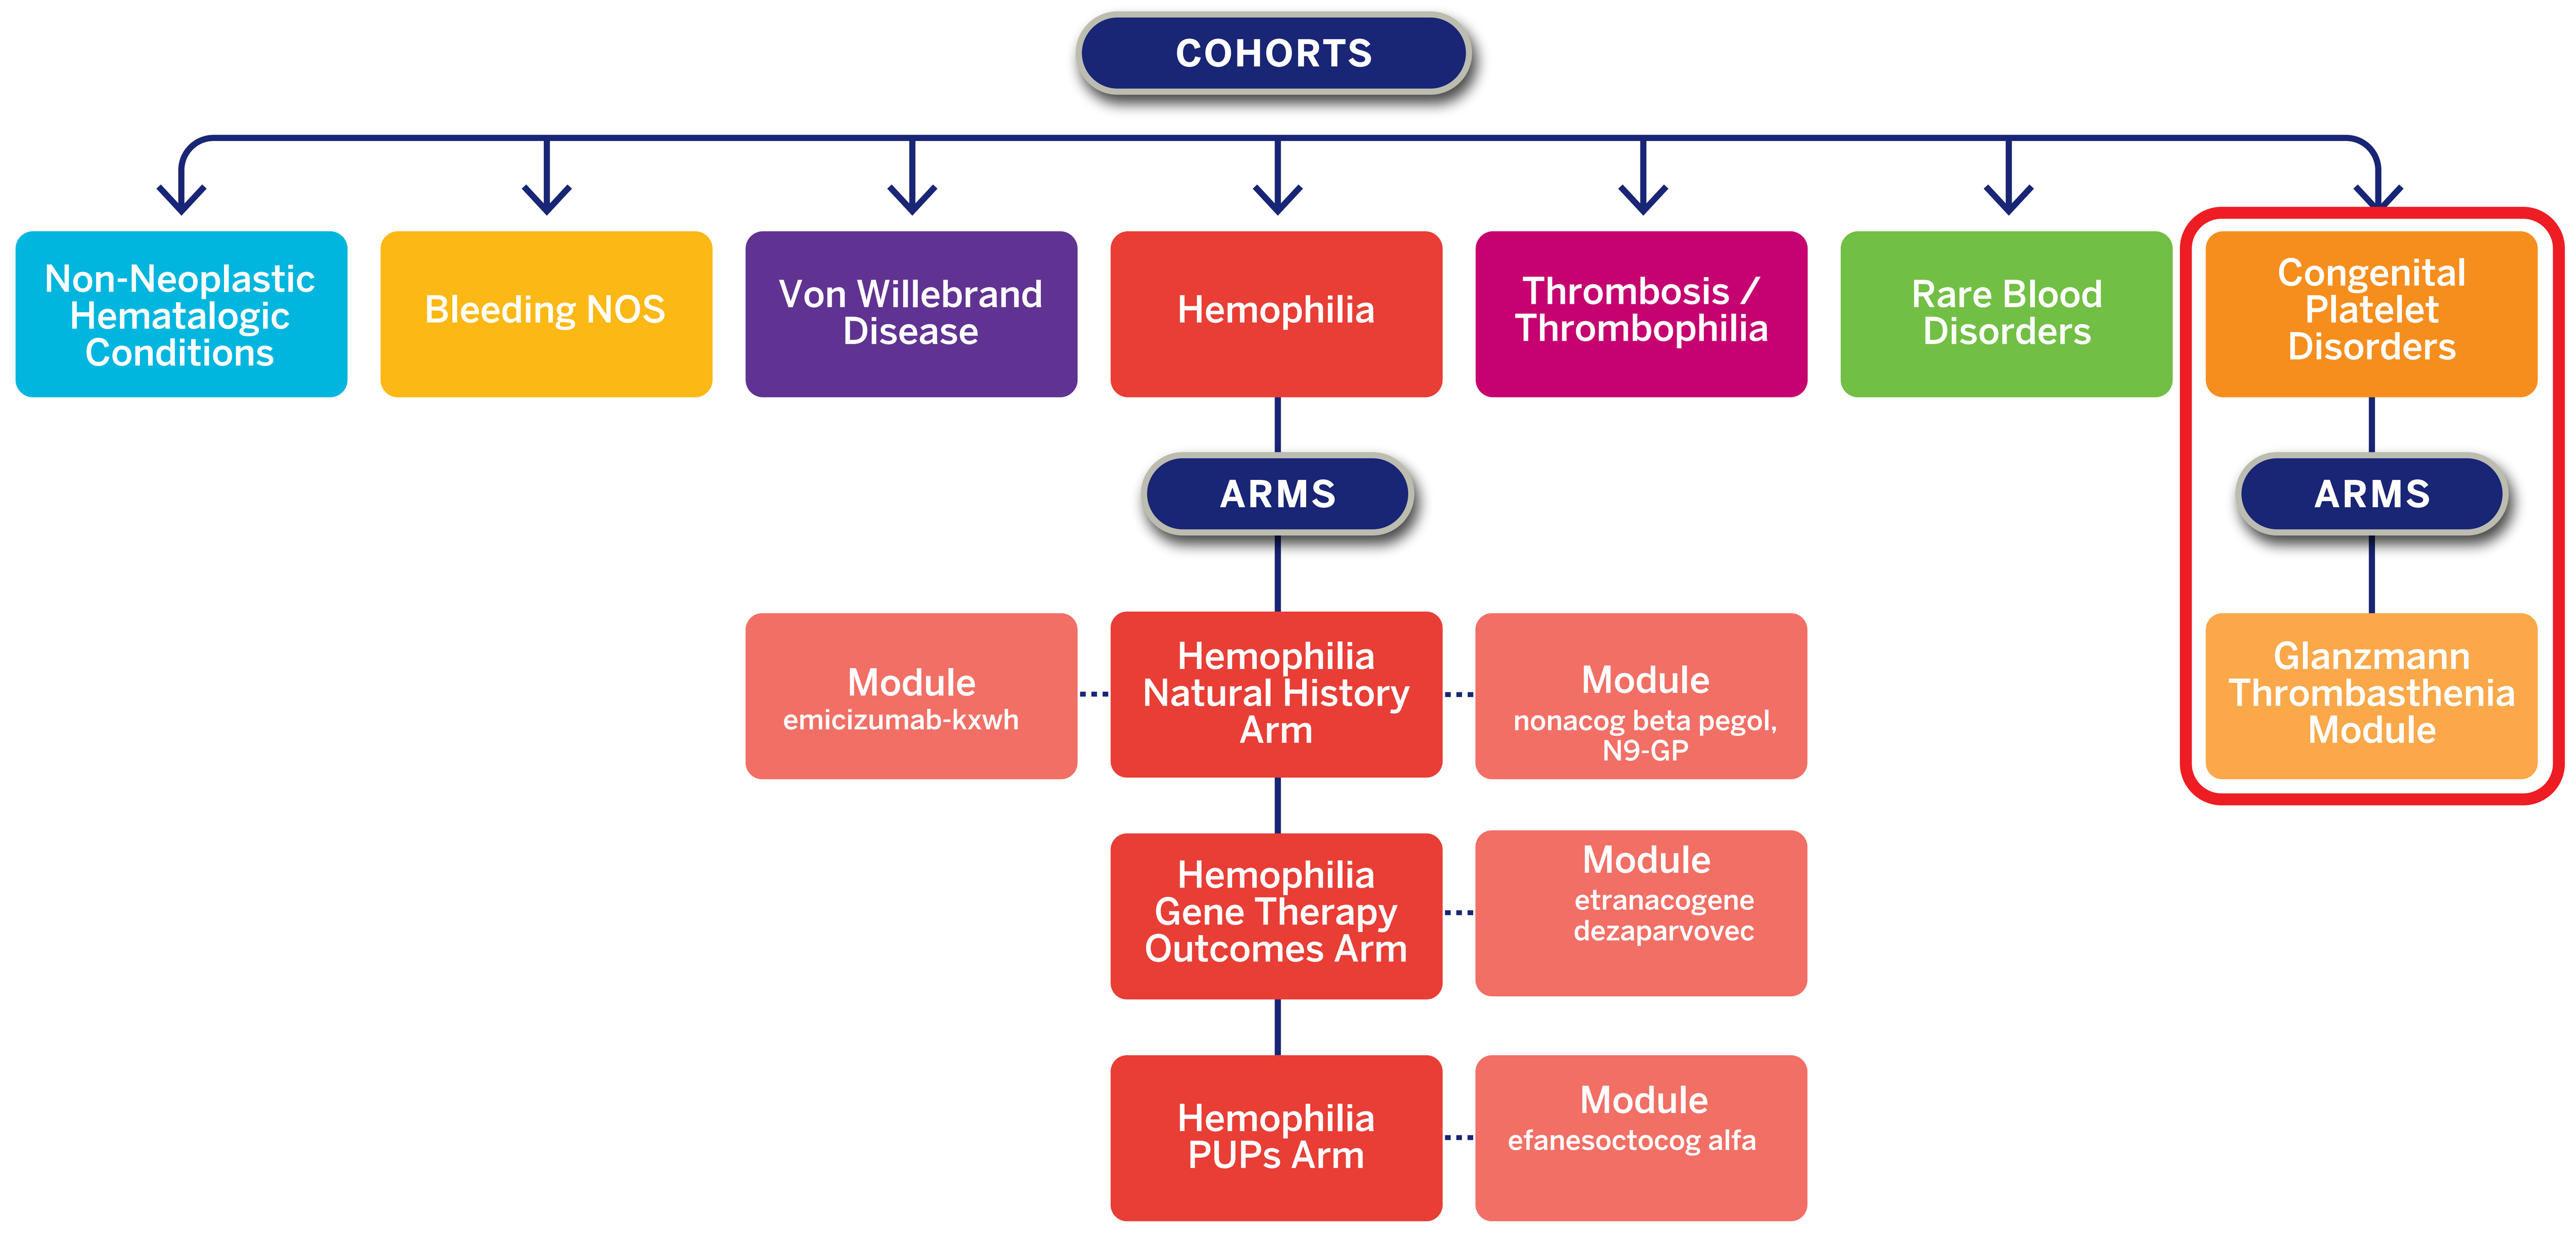
COHORTS
Non-Neoplastic Hematologic Conditions
Bleeding NOS
Von Willebrand Disease
Hemophilia
Thrombosis / Thrombophilia
Rare Blood Disorders
Congenital Platelet Disorders
ARMS
Module emicizumab-kxwh
Hemophilia Natural History Arm
Module nonacog beta pegol, N9-GP
Hemophilia Gene Therapy Outcomes Arm
Module etranacogene dezaparvovec
Hemophilia PUPs Arm
Module efanesoctocog alfa
ARMS
Glanzmann Thrombasthenia Module

Objectives of the Glanzmann Thrombasthenia Module

To describe the bleeding phenotype in GT including:

- Evaluating:
 - Frequency of bleeds
 - Patient-reported outcomes
 - Healthcare and treatment utilization
- Frequency of bleeds
- Site of bleeds
- Real-world effectiveness of GT therapies
- Creating a database for future research

Methods

We will be collecting data around the actual bleeding rate in those who live with GT as well as more fully describing the lived experience of being impacted by GT. The study will be conducted through ATHN-affiliated centers caring for those impacted by GT.

Study Design

This study is a longitudinal, natural history, observational cohort study. Approximately 50 participants with GT will be enrolled at ATHN affiliated treatment centers in the United States and one international ATHN Affiliate. The minimum planned duration of study participation for participants is approximately 18 months. Pediatric and adult subjects with GT are expected to be enrolled. Each participant will complete a daily record for 3 months with treatment details, in addition to the lived experience of being impacted by GT.

Summary

ATHN Transcends has received central IRB approval and is currently being rolled out across participating ATHN Affiliates in the United States. Enrollment in the GT Module can begin as soon as a site opens ATHN Transcends to enrollment.

Conclusion

The GT Module of ATHN Transcends will allow collection of data that will help gather a better understanding of the phenotype and treatment efficacy of the various treatment modalities, which will facilitate creation of better treatment guidelines for people living with GT.

Poster # 110