Agreement Between a Chromogenic Modified Nijmegen-Bethesda Assay and a Qualitative ELISA test in Detection of Factor VIII Inhibitors in Plasma from Persons with Hemophilia A (PwHA)

Ali Sadeghi-Khomami, Marc Boylan: Precision BioLogic Inc., Dartmouth, NS, Canada
David C. Chen, Joanne I. Adamkewicz: Genentech Inc., South San Francisco, CA, USA

Presented at WFH 2018 World Congress, May 20-24, Glasgow, Scotland

PUSHED FOR TIME? In order to use the QR code, please use or download an app called 'QR code reader' from the Apple Appstore or the Android Playstore
Receive an instant copy of this poster
Request additional posters and presentations of trials sponsored/supported by Roche at this congress
If you are having difficulties using the QR code reader, please visit: http://bit.ly/2E1sHES

PrecisionBioLogic

Background/Objectives

- An inhibitor response via antibody development to Factor VIII (FVIII) replacement therapies is the most significant complication of hemophilia A treatment today.
- Anti-FVIII antibodies can block a functional area of FVIII (inhibitory) or other sites of FVIII molecule (non-inhibitory).
- Immucor FVIII Antibody Screen, a solid-phase indirect FVIII ELISA, is a qualitative assay designed to detect IgG antibodies against a full-length recombinant human FVIII (plate coated with Kogenate FS). False-positive ELISA results may occur with lupus anticoagulant and anti-phospholipid antibodies.[1] Thus, results need confirmation by a more specific assay.
- An accurate diagnosis of FVIII inhibitors is essential in guiding patient management, however quantification of inhibitor titers by clot-based functional assays revealed coefficients of variation as high as 50% between laboratories in international proficiency studies.[2] Different reagents and methods used across labs (e.g. plasma sources, absence of or variations in heat deactivation procedures, use of buffered vs. non-buffered plasma) account for the high variability.
- Chromogenic endpoint assays have better specificity than one-stage clot-based assays since the latter depend on fibrin clot formation which is impacted by the presence of heparin, lupus anticoagulants and inhibitors of coagulation factors other than FVIII.[3]
- Thus, chromogenic Modified Nijmegen-Bethesda Assay (MNBA) has potential for standardization and improvement of the FVIII inhibitor assay.

Conclusions

- In this study, we found Immucor anti-FVIII ELISA to be a sensitive assay in the detection of anti-FVIII antibodies, suitable for batch screening of anti-FVIII antibodies and requiring only small plasma volumes.
- A confirmatory inhibitory assay, such as chromogenic MNBA, is required after ELISA screening for detection of inhibitor antibodies and titer quantification.
- The MNBA kit shows promise for laboratories seeking a standardized chromogenic FVIII inhibitor assay suitable for clinical management or multi-center clinical studies of PwHA.

References

[1] Sahud M. *et al*. False-positive results in ELISA-based anti-FVIII antibody assay occur with lupus anticoagulant and phospholipid antibodies. Haemophilia 2012; 18:771-781.

[2] Favaloro E.J. *et al*. Laboratory testing for factor inhibitors. Haemophilia 2014; 20(S4):94-98.

[3] Moser K.A. *et al*. Chromogenic factor VIII activity assay. Am J Hematol 2014; 89(7):781-783.

[4] Boylan B. *et al*. Effects of pre-analytical heat treatment in factor VIII (FVIII) inhibitor assays on FVIII antibody levels. Hemophilia. 2018 Feb 20. doi: 10.1111/hae.13435.

[5] Miller C.H. *et al*. Validation of Nijmegen-Bethesda assay modifications to allow inhibitor measurement during replacement therapy and facilitate inhibitor surveillance. Thromb Haemostasis 2012; 10:1055-1061.

[6] Batty P. *et al*. Pre-analytical heat treatment and a FVIII ELISA improve Factor VIII antibody detection in acquired haemophilia A. Br J Haematol 2014; 166:953-956.

[7] Blanchette V.S. *et al*. Definitions in hemophilia: communication from the SSC of the ISTH. Thromb Haemostasis 2014; 12:1935-1939.

Acknowledgements: This study was funded by F. Hoffmann-La Roche Ltd. Editorial assistance was provided by Laurie Ketch and Karen Black (Precision BioLogic Inc.).

Disclosures: Sadeghi-Khomami A and Boylan M are employees of Precision BioLogic Inc. Adamkewicz JI and Chen DC are employees of Genentech Inc. and stock shareholders of Roche.

Methods

- To eliminate FVIII depleted plasma as a potential source of variation and to standardize inhibitor titer measurement, a kit for MNBA was developed with these components:
 - IB-PNP: Imidazole Buffered Pooled Normal Plasma (pH = 7.4, 100 mM Imidazole, FVIII 95-105%) to provide a source of native FVIII and prevent pH change during incubation.
 - IB-BSA: Imidazole Buffered Bovine Serum Albumin (pH = 7.4, 4% w/v BSA in 50 mM Imidazole) to replace FVIII depleted plasma in the Nijmegen assay.
 - POS-Ctrl: Positive FVIII inhibitor control (~1 BU/mL, polyclonal anti-human FVIII antibody in a buffered human FVIII-depleted plasma).
 - NEG-Ctrl: FVIII inhibitor-free human plasma (buffered).
- The MNBA kit components were frozen and stored at < -70 °C until use **(Figure 1)**.
- A total of 37 frozen plasma samples from PwHA as well as 33 frozen plasma samples from normal donors were thawed, heat deactivated, and centrifuged. The supernatant was drawn off and stored at < -70 °C until testing with chromogenic MNBA and Immucor ELISA **(Figure 2)**.
- A heat deactivation step was incorporated in sample preparation to dissociate antibody-FVIII complexes and to eliminate remaining FVIII activity in plasma samples from PwHA, thus preventing the likelihood of false negative results.[4,5,6]
- Chromogenic MNBA: after thawing the heat deactivated plasma samples and FVIII Inhibitor Kit controls (Test Samples), a 1:1 mixture of IB-PNP, with either undiluted or IB-BSA pre-diluted Test Samples, were prepared (Test Mix, 400 µL).
- A 1:1 Control Mix was prepared with IB-PNP:IB-BSA (400 µL).
- The Control Mix and Test Mixes were incubated for 2h at 37 °C in a water bath followed by a 10 min. incubation on ice.
- After incubation, FVIII activities in the mixed samples were determined on a Siemens BCS® XP analyzer using Siemens Factor VIII Chromogenic Assay.
- The Test Mix dilution with residual FVIII activity closest to 50% and within the accepted range of 25-75% was used to calculate the FVIII inhibitor titer in Bethesda Units **(Figure 3)**.
- Anti-FVIII ELISA: after thawing, the heat deactivated plasma samples and kit-provided serum controls were diluted 1:4 with TRIS-BSA-Saline, and 15 µL of each were tested in duplicate according to the manufacturer's instructions.
- Anti-FVIII antibodies in samples were detected by the anti-human goat IgG conjugated to alkaline phosphatase and *p*-nitrophenol phosphate (PNPP) substrate supplied by the kit. The optical density (OD) of the color development was measured at 405 nm using a SoftMax plate reader.

Results

- All normal plasma samples were negative on both assays **(Figure 4, Figure 5)**.
- Results for 33 out of 37 plasma samples from PwHA were concordant between ELISA and chromogenic MNBA **(Table 1)**.
- Four discrepant results were borderline positive by anti-FVIII ELISA method but did not show FVIII inhibitory effect by chromogenic MNBA **(Figure 4, Figure 5)**.
- Further analysis of the four discrepant samples was performed by testing with a clot-based MNBA. The clot-based MNBA result was concordant with the chromogenic MNBA result (i.e. negative) in all four cases (data not shown).

Figure 4
ELISA results

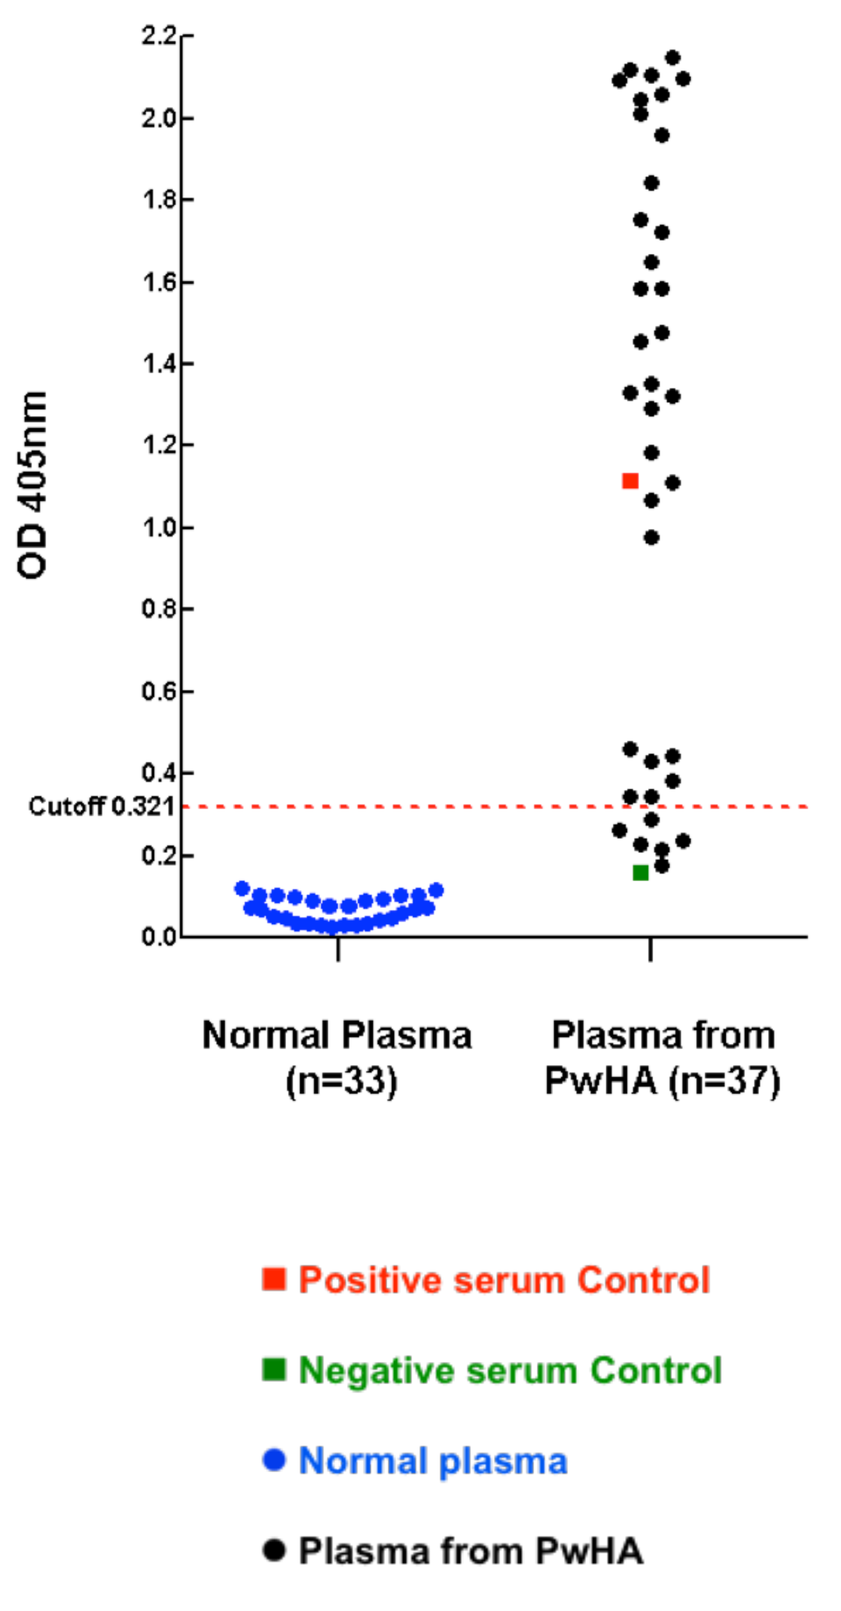
OD 405nm
Cutoff 0.321
Normal Plasma (n=33)
Plasma from PwHA (n=37)
Positive serum Control
Negative serum Control
Normal plasma
Plasma from PwHA

Figure 5
Chromogenic MNBA results

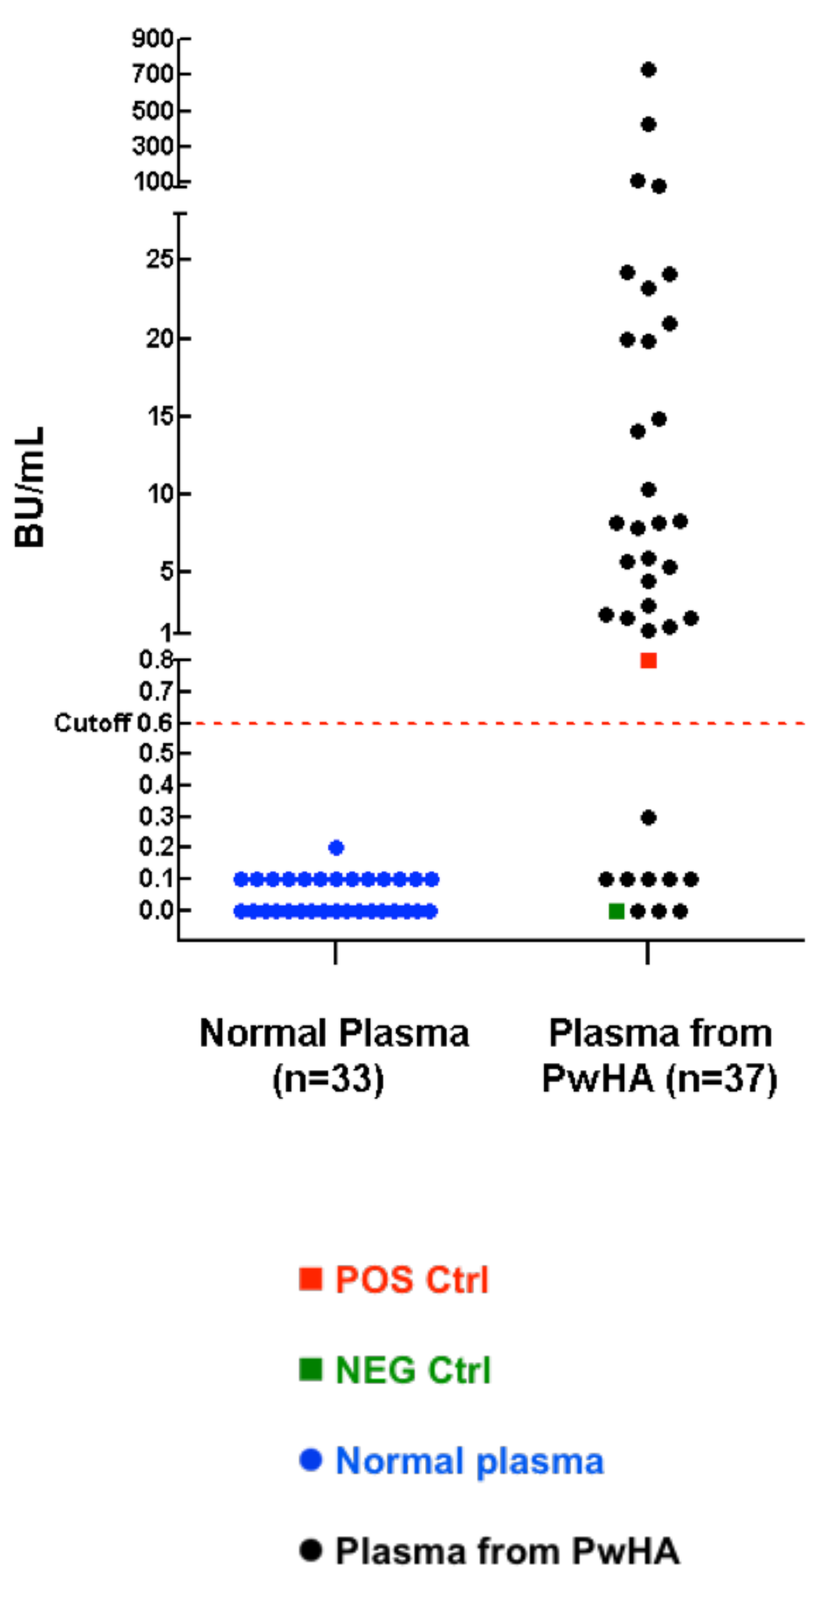
BU/mL
Cutoff 0.6
Normal Plasma (n=33)
Plasma from PwHA (n=37)
POS Ctrl
NEG Ctrl
Normal plasma
Plasma from PwHA

Table 1
Diagnostic agreement between chromogenic MNBA* and ELISA**

FVIII Inhibitor detection in plasma samples from PwHA (n=37)		Immucor anti-FVIII ELISA	
		Positive	Negative
Chromogenic MNBA	Positive	27	0
	Negative	4	6
Agreement		Percent	95% CI
Positive Percent Agreement (PPA)		87.1%	70-96%
Negative Percent Agreement (NPA)		100%	54-100%
Total Percent Agreement (TPA)		89.2%	75-97%

* ≥0.6 BU/mL cutoff (a consensus recommendation by ISTH-SSC)[7]
** >0.321 OD_{405nm} cutoff (determined by a lot specific cutoff control provided in ELISA kit)

Figure 1

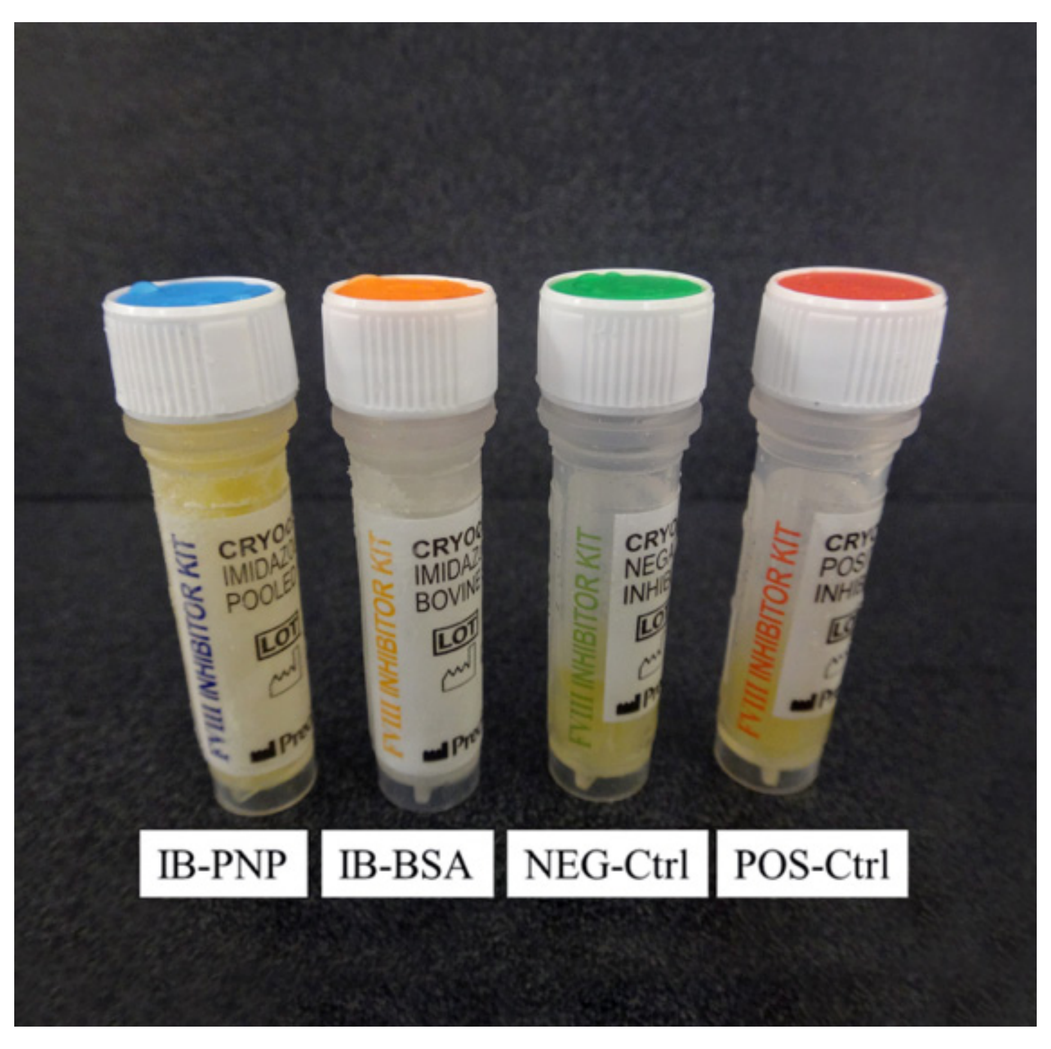
IB-PNP
IB-BSA
NEG-Ctrl
POS-Ctrl

Figure 2

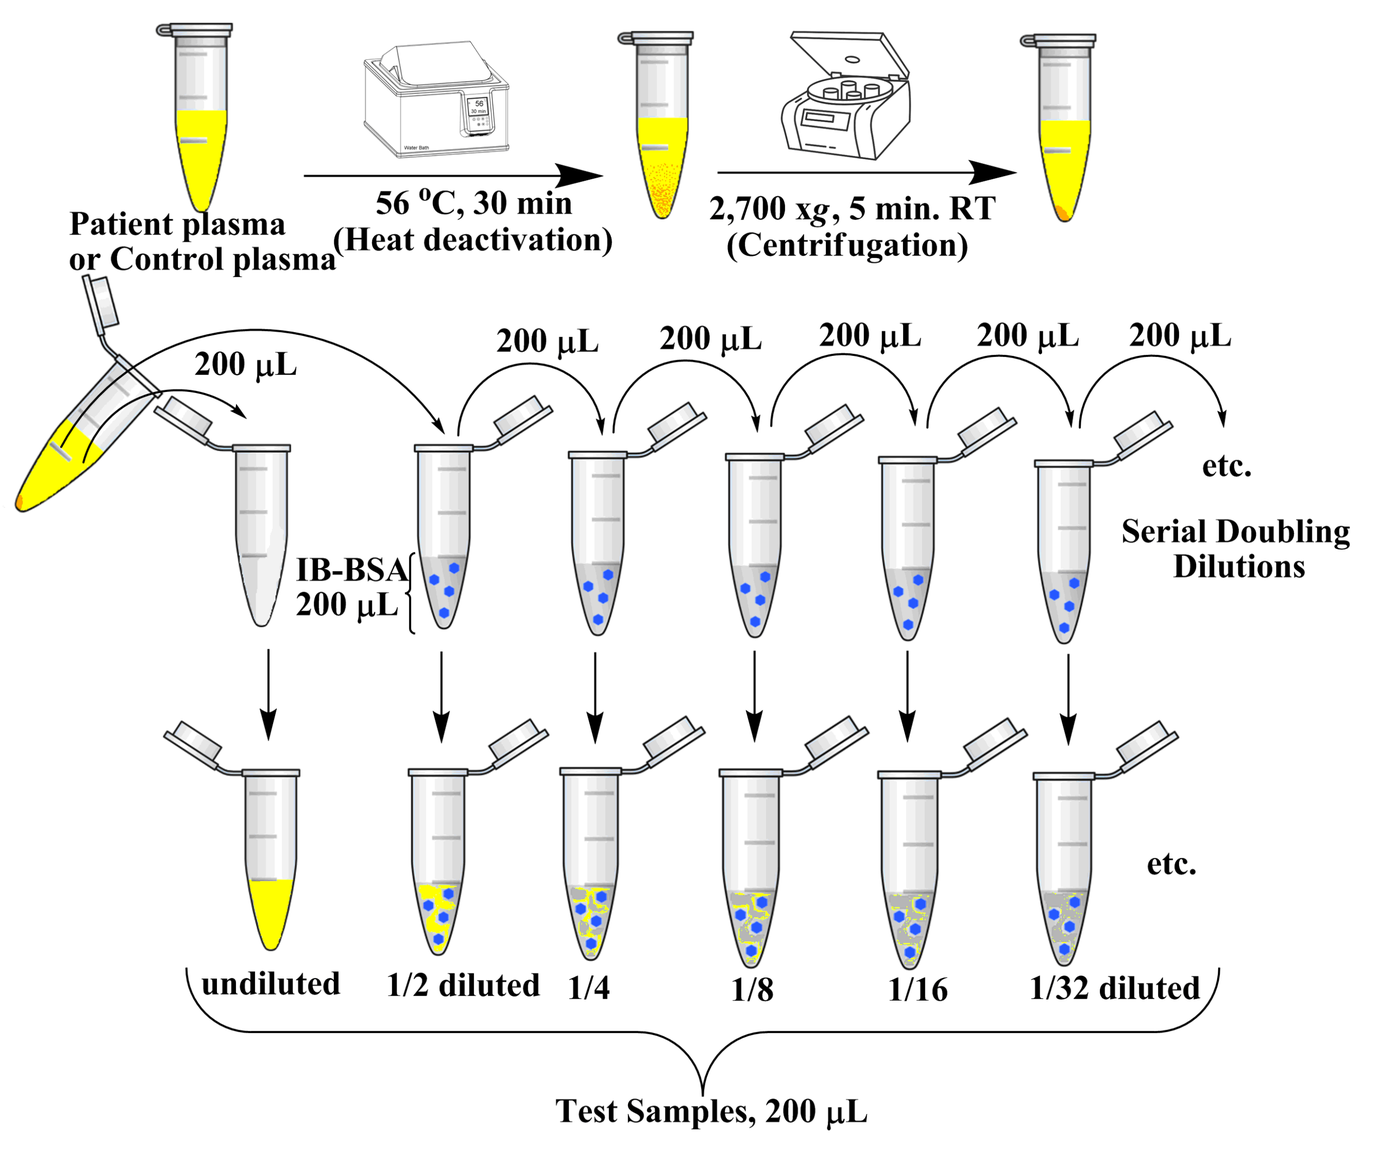
Patient plasma or Control plasma
56 °C, 30 min (Heat deactivation)
2,700 xg, 5 min. RT (Centrifugation)
200 µL
IB-BSA 200 µL
Serial Doubling Dilutions
etc.
undiluted
1/2 diluted
1/4
1/8
1/16
1/32 diluted
Test Samples, 200 µL

Figure 3

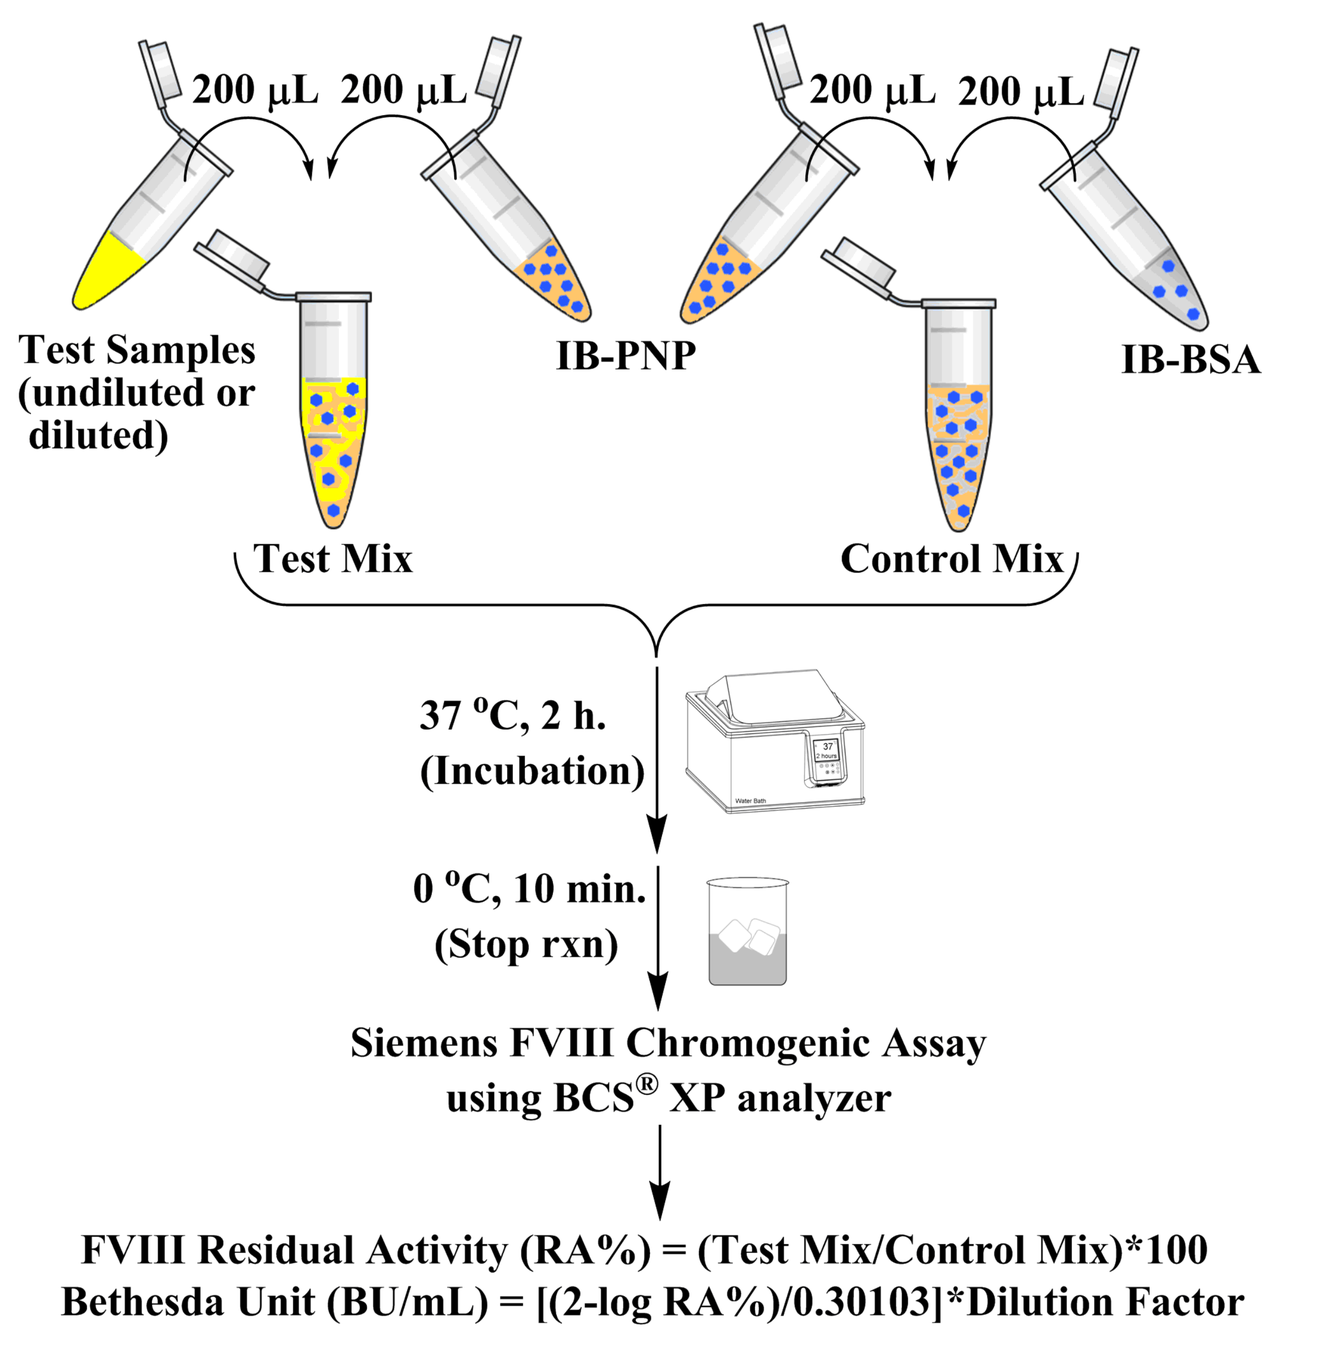
200 µL 200 µL
Test Samples (undiluted or diluted)
IB-PNP
IB-BSA
Test Mix
Control Mix
37 °C, 2 h. (Incubation)
0 °C, 10 min. (Stop rxn)
Siemens FVIII Chromogenic Assay using BCS® XP analyzer
FVIII Residual Activity (RA%) = (Test Mix/Control Mix)*100
Bethesda Unit (BU/mL) = [(2-log RA%)/0.30103]*Dilution Factor

WFH2018 M-P-59
Inhibitors, management of
Ali Sadeghi-Khomami
DOI: 10.3252/pso.eu.WFH2018.2018
Poster presented at: WFH 2018 World Congress Glasgow, Scotland May 20-24
PosterSessionOnline.com