Poster 89-PP-M

Interaction of BAX 826 (PSAylated rFVIII) With VWF and LRP1 – An *in vitro* and *in vivo* Assessment

Schrenk G, Kopić A, Knappe S, Billwein M, Schaedler M, Turecek PL, Hoellriegl W, Dockal M, Scheiflinger F

Shire, Vienna, Austria

INTRODUCTION

- BAX 826, Baxalta's second investigational extended half-life candidate is based on ADVATE [antihemophilic factor (recombinant)], a full length recombinant FVIII molecule with an established extensive safety and efficacy profile, and is modified using a hydrophilic polymer, polysialic acid (PSA), to extend dosing intervals.
- Extended FVIII circulation times would reduce the frequency of infusions, increase patient compliance, reduce the number of bleeds, and allow higher trough levels of FVIII to be reached.
- Most FVIII molecules in the circulation are complexed with von Willebrand factor (VWF), which prevents binding of FVIII to LRP1 (low density lipoprotein receptor-related protein 1), an important receptor involved in the clearance of FVIII. While the half-life of free FVIII is 2-3 h, that of FVIII bound to VWF is approximately 17 h, demonstrating that VWF is a major determinant of FVIII circulation time.

OBJECTIVE

- To investigate the interaction of BAX 826 with von VWF and LRP1 using appropriate *in vitro* methods.
- To compare the VWF-independent clearance between BAX 826 and rFVIII in mice deficient in FVIII and VWF.

METHODS

- The kinetics of FVIII binding to LRP1 and to VWF were determined using surface plasmon resonance technology (Biacore, GE Healthcare, Uppsala, Sweden):
 - LRP1 (BioMac, Leipzig, Germany) was immobilized on the flow cells of a CM4 biosensor chip at a density of 5000 response units (RU). A series of dilutions (21 to 357 nM) of rFVIII or BAX 826 were then applied to the chip, allowing 10 min for association and 5 min for dissociation. The equilibrium dissociation constant (KD) was determined assuming a 1:1 interaction.
 - Plasma-derived VWF (pdVWF, Diagnostica Stago, Asnières sur Seine, France) was immobilized at three densities (300, 600, and 1200 response units (RU)) on the flow cells of a CM5 biosensor chip. A series of dilutions (0.18 to 5 nM) of rFVIII or BAX 826 were then applied to the chip, allowing 10 min for association and 5 min for dissociation. The equilibrium dissociation constant (KD) was determined assuming a 1:1 interaction.
- ELISA combined chromogenic assays were used as an orthogonal methods to the Biacore assays to study FVIII-LRP1 and FVIII-VWF binding. In brief,
 - A dilution series (0 to 10 IU/mL FVIII activity) of rFVIII or BAX 826 was added to the wells of a microtiter plate coated with LRP1. After incubation and removal of unbound FVIII by a washing step, bound FVIII was quantified by a FVIII chromogenic assay (Technochrom FVIII:C kit, Technoclone, Vienna, Austria).
 - A polyclonal anti-human VWF antibody was immobilized on the wells of a microtiter plate followed by incubation with 0.1 IU/mL VWF:Ag of VWF (recombinant VWF, Baxalta, Vienna , Austria). Subsequently, a series of dilutions (0.016 to 1 IU/mL FVIII activity) of rFVIII or BAX 826 were added. After incubation and removal of unbound FVIII by a washing step, VWF-bound FVIII was quantified by FVIII chromogenic assay.
- The pharmacokinetics of rFVIII and BAX 826 were evaluated in a VWFxFVIII double knockout mouse model. After intravenous administration of 200 IU/kg rFVIII, blood samples were collected by cardiac puncture at several time points from 5 min to 7 h post injection. FVIII levels in plasma stabilized by *ex vivo* addition of 1 IU/mL VWF were determined by a chromogenic activity assay. The endpoints for statistical evaluation were terminal half-life and mean residence time (MRT).

RESULTS – *in vitro* Binding Studies

Interaction of rFVIII and BAX 826 With LRP1 Clearance Receptor

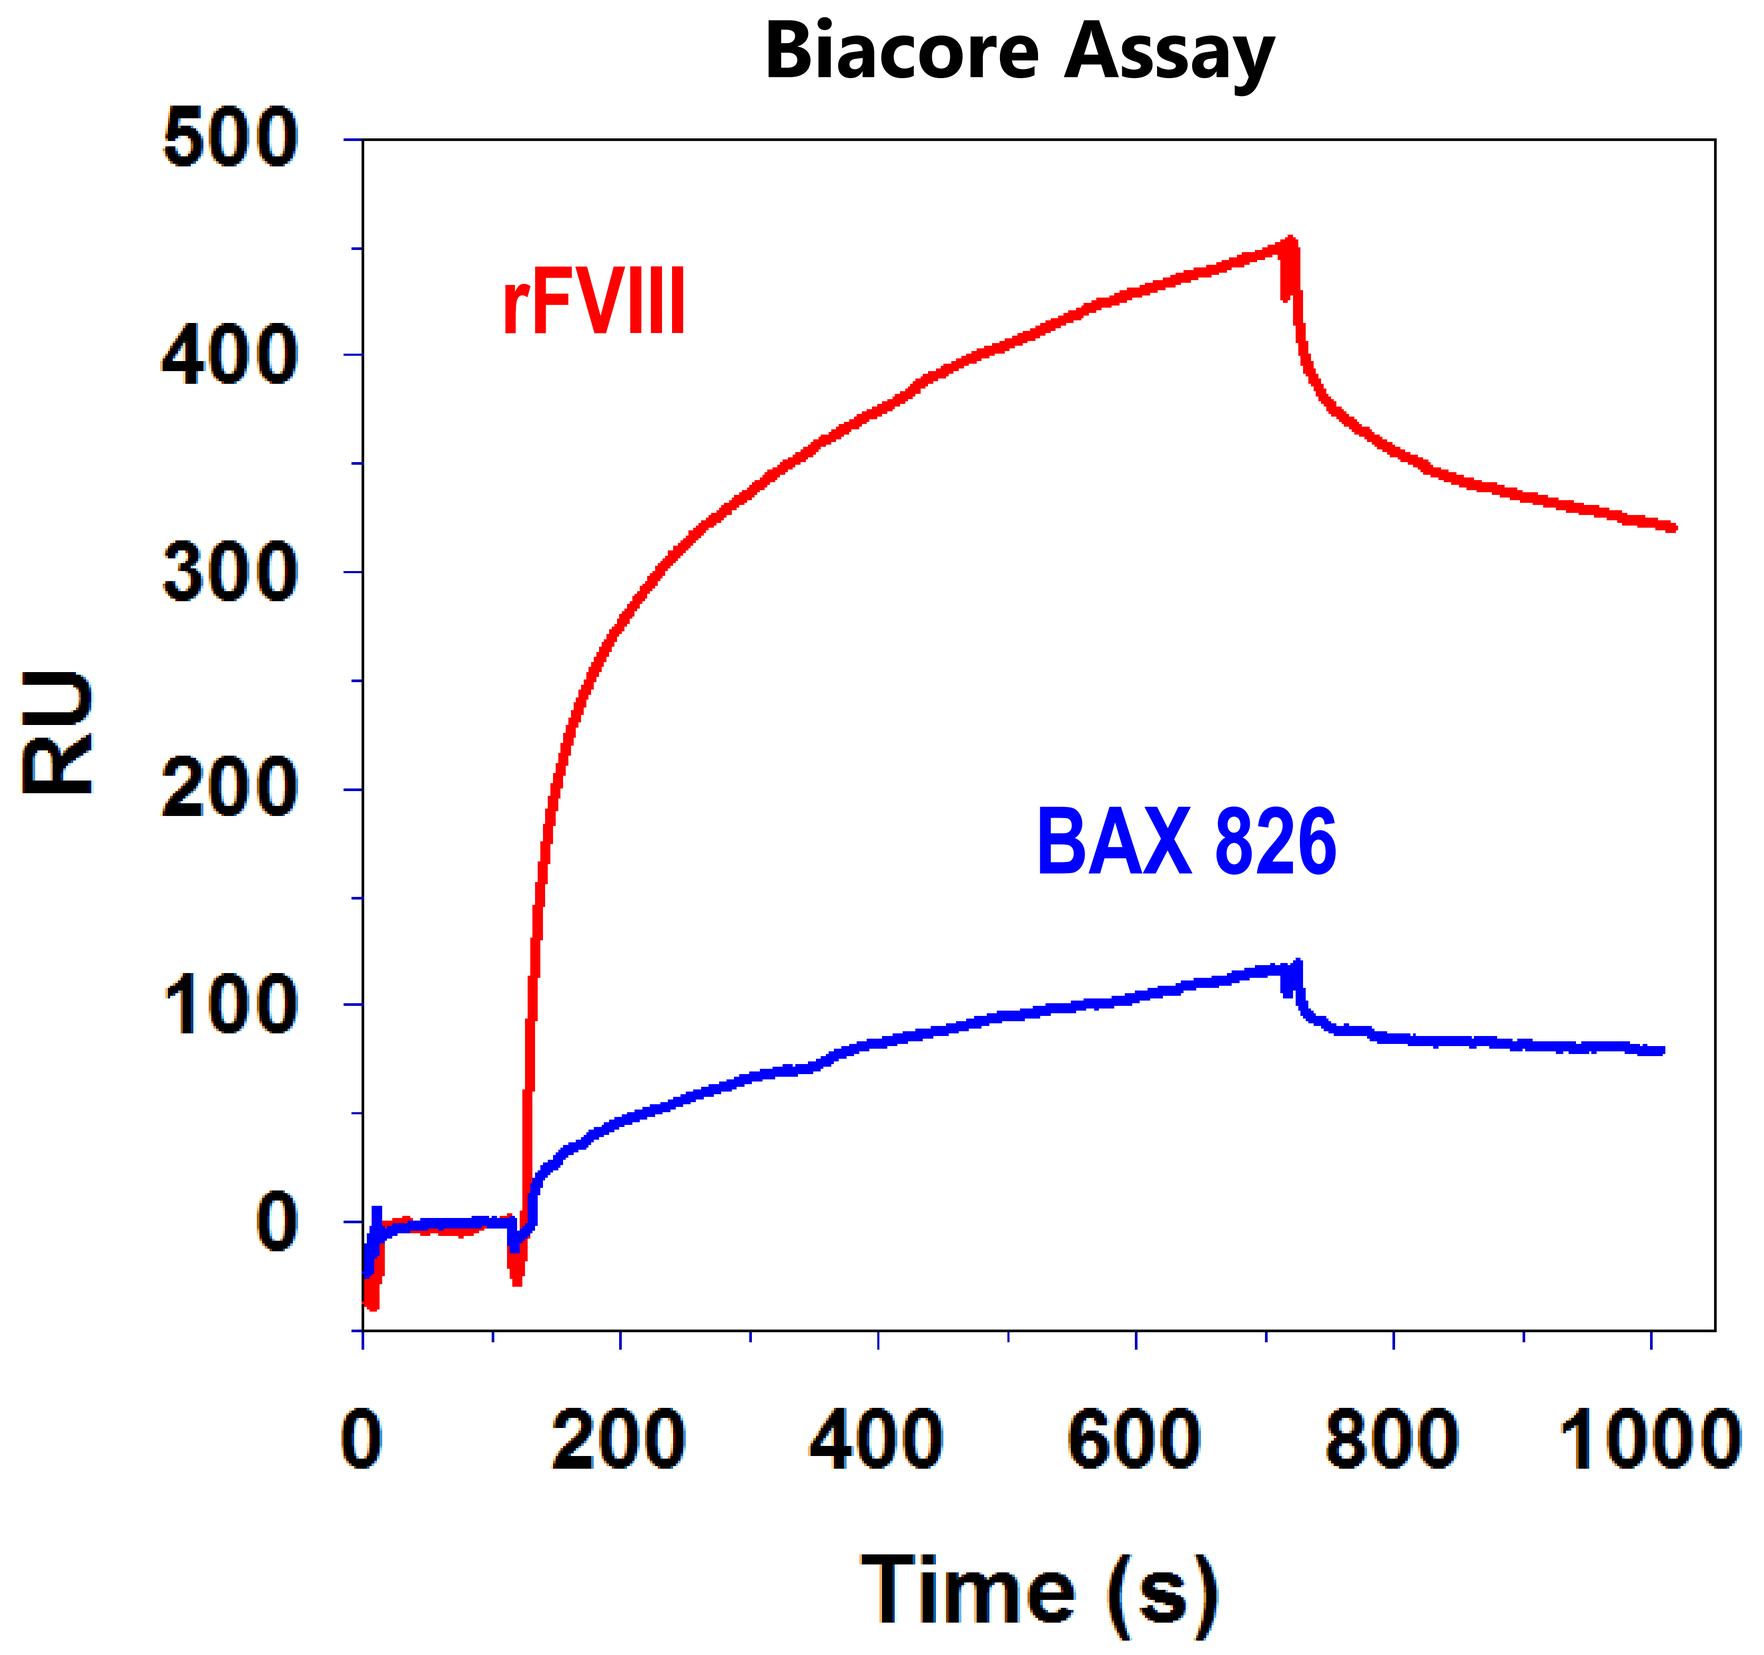
Biacore Assay
RU
500
400
300
200
100
0
rFVIII
BAX 826
0
200
400
600
800
1000
Time (s)

Figure 1: Representative example of a sensorgram of the binding of 100 µg/mL rFVIII or BAX 826 to immobilized LRP1.

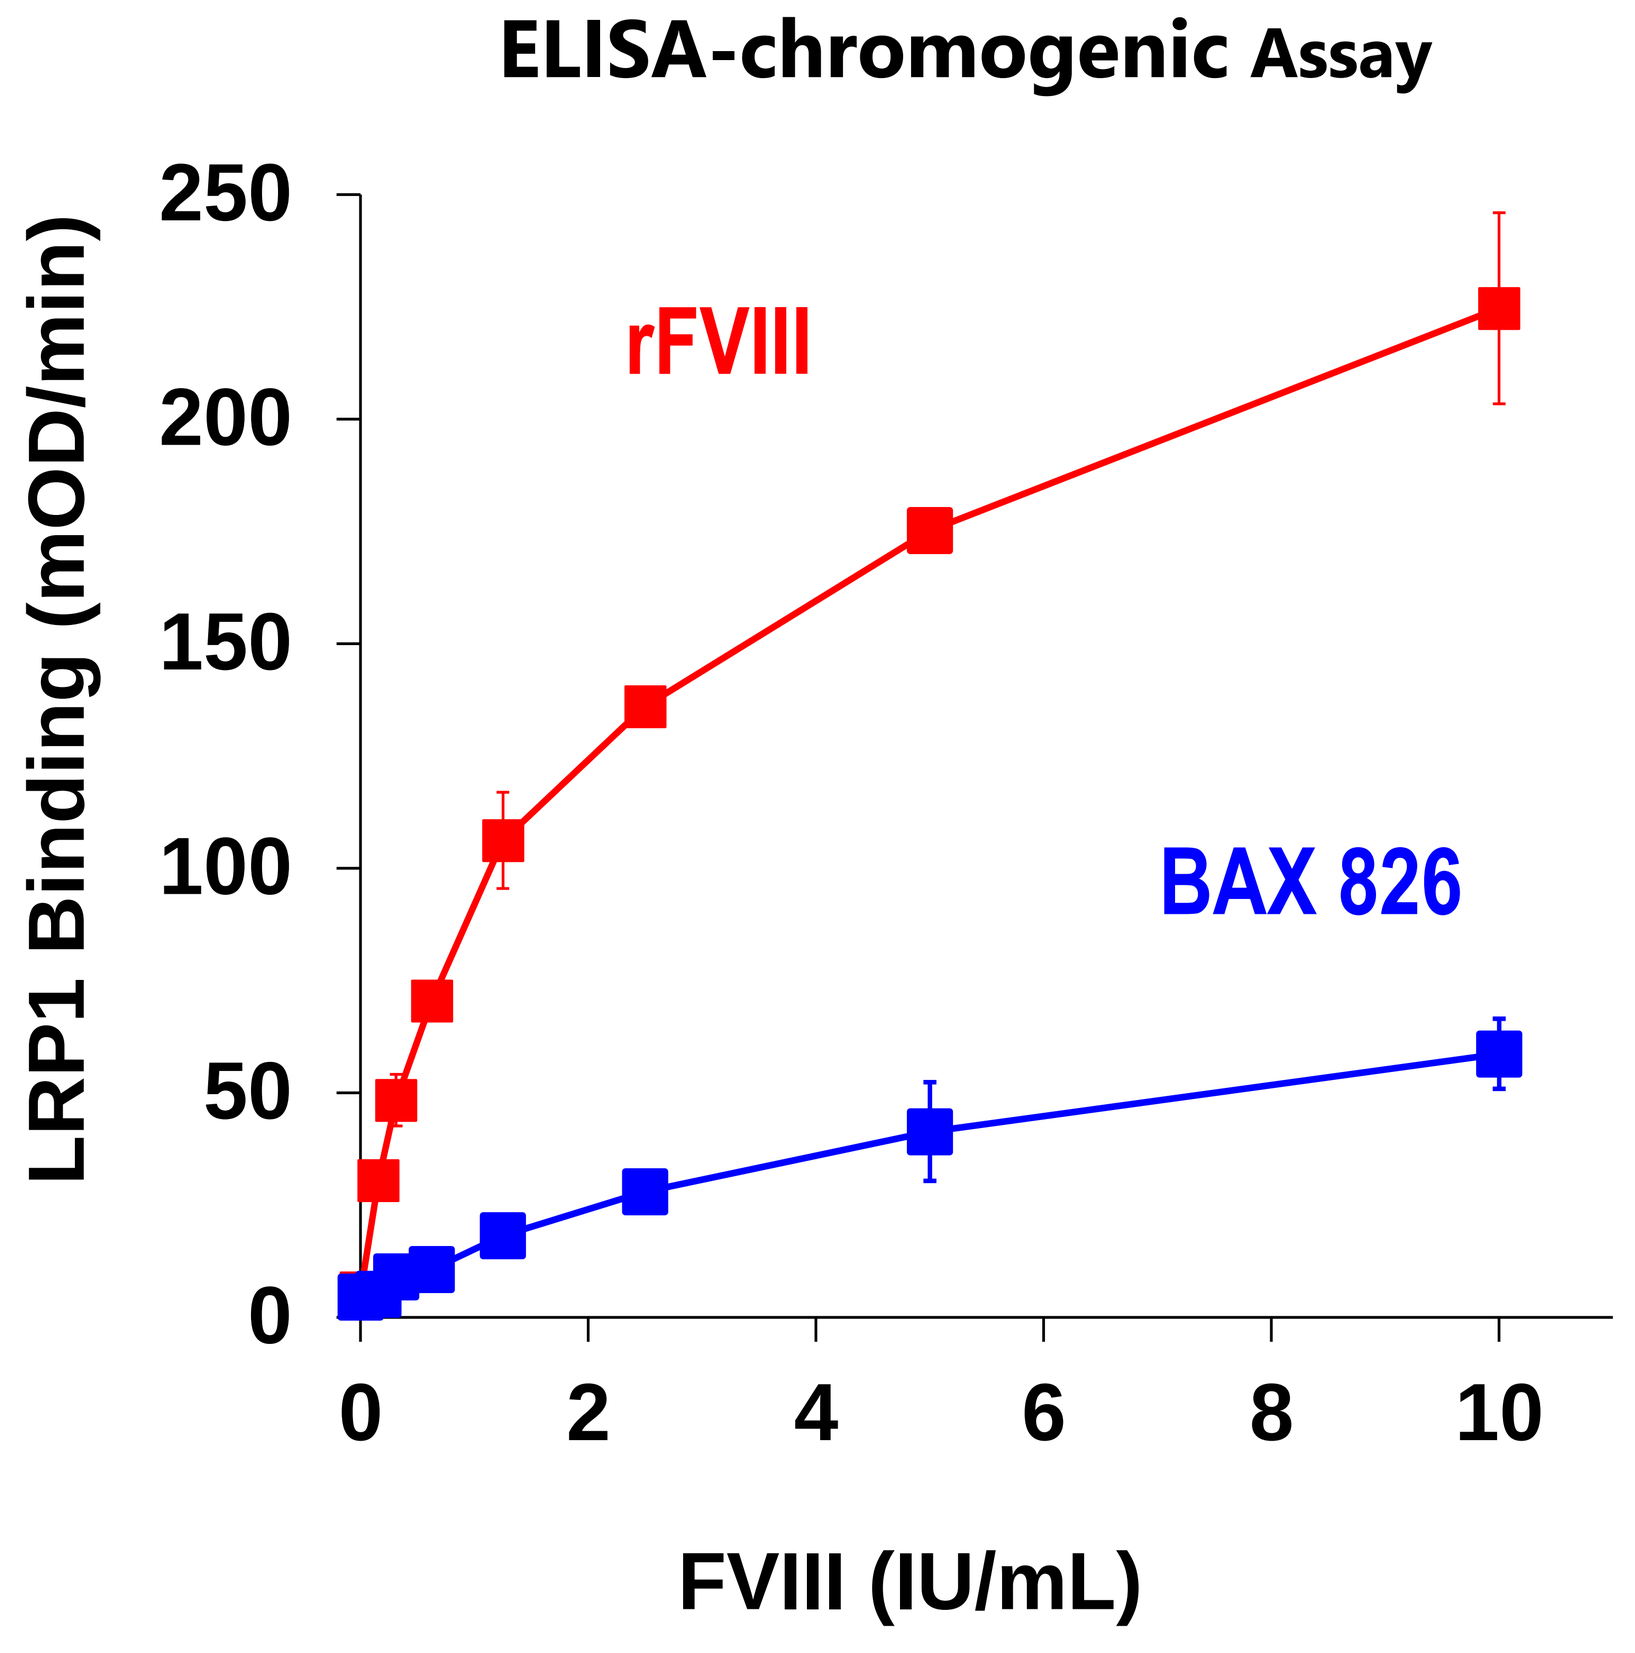
ELISA-chromogenic Assay
LRP1 Binding (mOD/min)
250
200
150
100
50
0
rFVIII
BAX 826
0
2
4
6
8
10
FVIII (IU/mL)

Figure 2: Results for three independent measurements. Error bars show standard deviation.

- Binding kinetics of rFVIII and BAX 826 to LRP1 were determined using a Biacore assay. The overall binding response of BAX 826 to LRP1 was markedly lower than to rFVIII. This reduced binding capacity is considered to be the result of rFVIII modification with PSA, which yields a rFVIII conjugate where specific binding epitopes for VWF are shielded by PSA.
- The binding affinities of BAX 826 and rFVIII were similar, with K_D values ranging from 12.6 to 41.0 nM for BAX 826 (n = 6) and from 18.6 to 28.0 nM for rFVIII (n = 4). These results indicate functional intact binding epitopes on BAX 826 for LRP1 when not shielded by PSA.
- Data from the ELISA-combined chromogenic assay for FVIII were in agreement with those from Biacore analysis, confirming reduced binding of BAX 826 to LRP1.

Interaction of rFVIII and BAX 826 With VWF

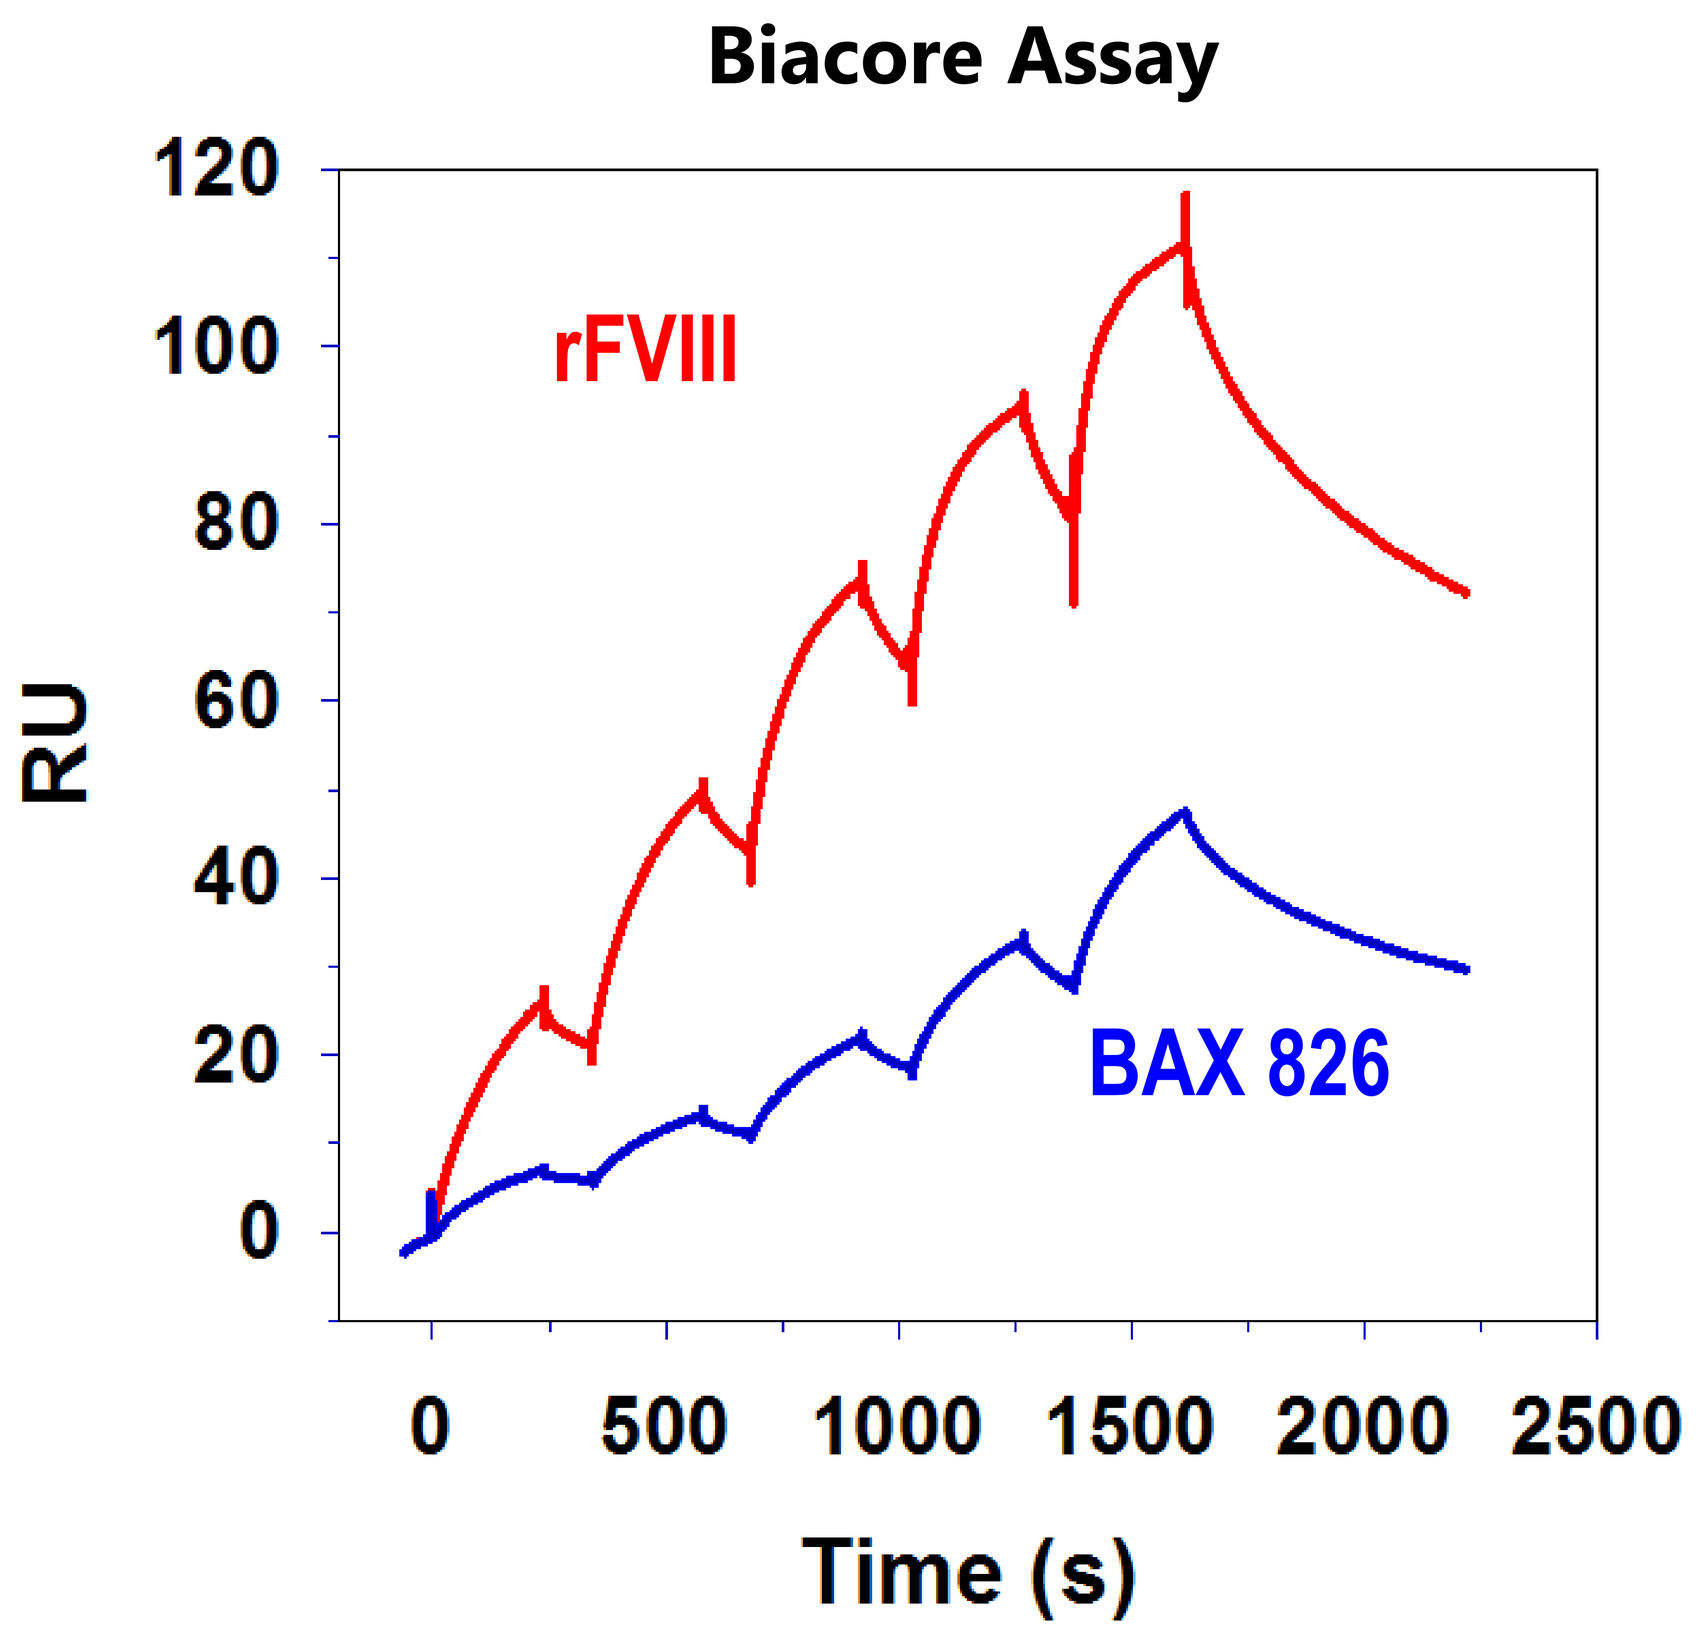
Biacore Assay
RU
120
100
80
60
40
20
0
rFVIII
BAX 826
0
500
1000
1500
2000
2500
Time (s)

Figure 3: Representative example of a sensorgram of the binding of 0.18 to 5 nM rFVIII or BAX 826 to immobilized VWF (1200 RU)

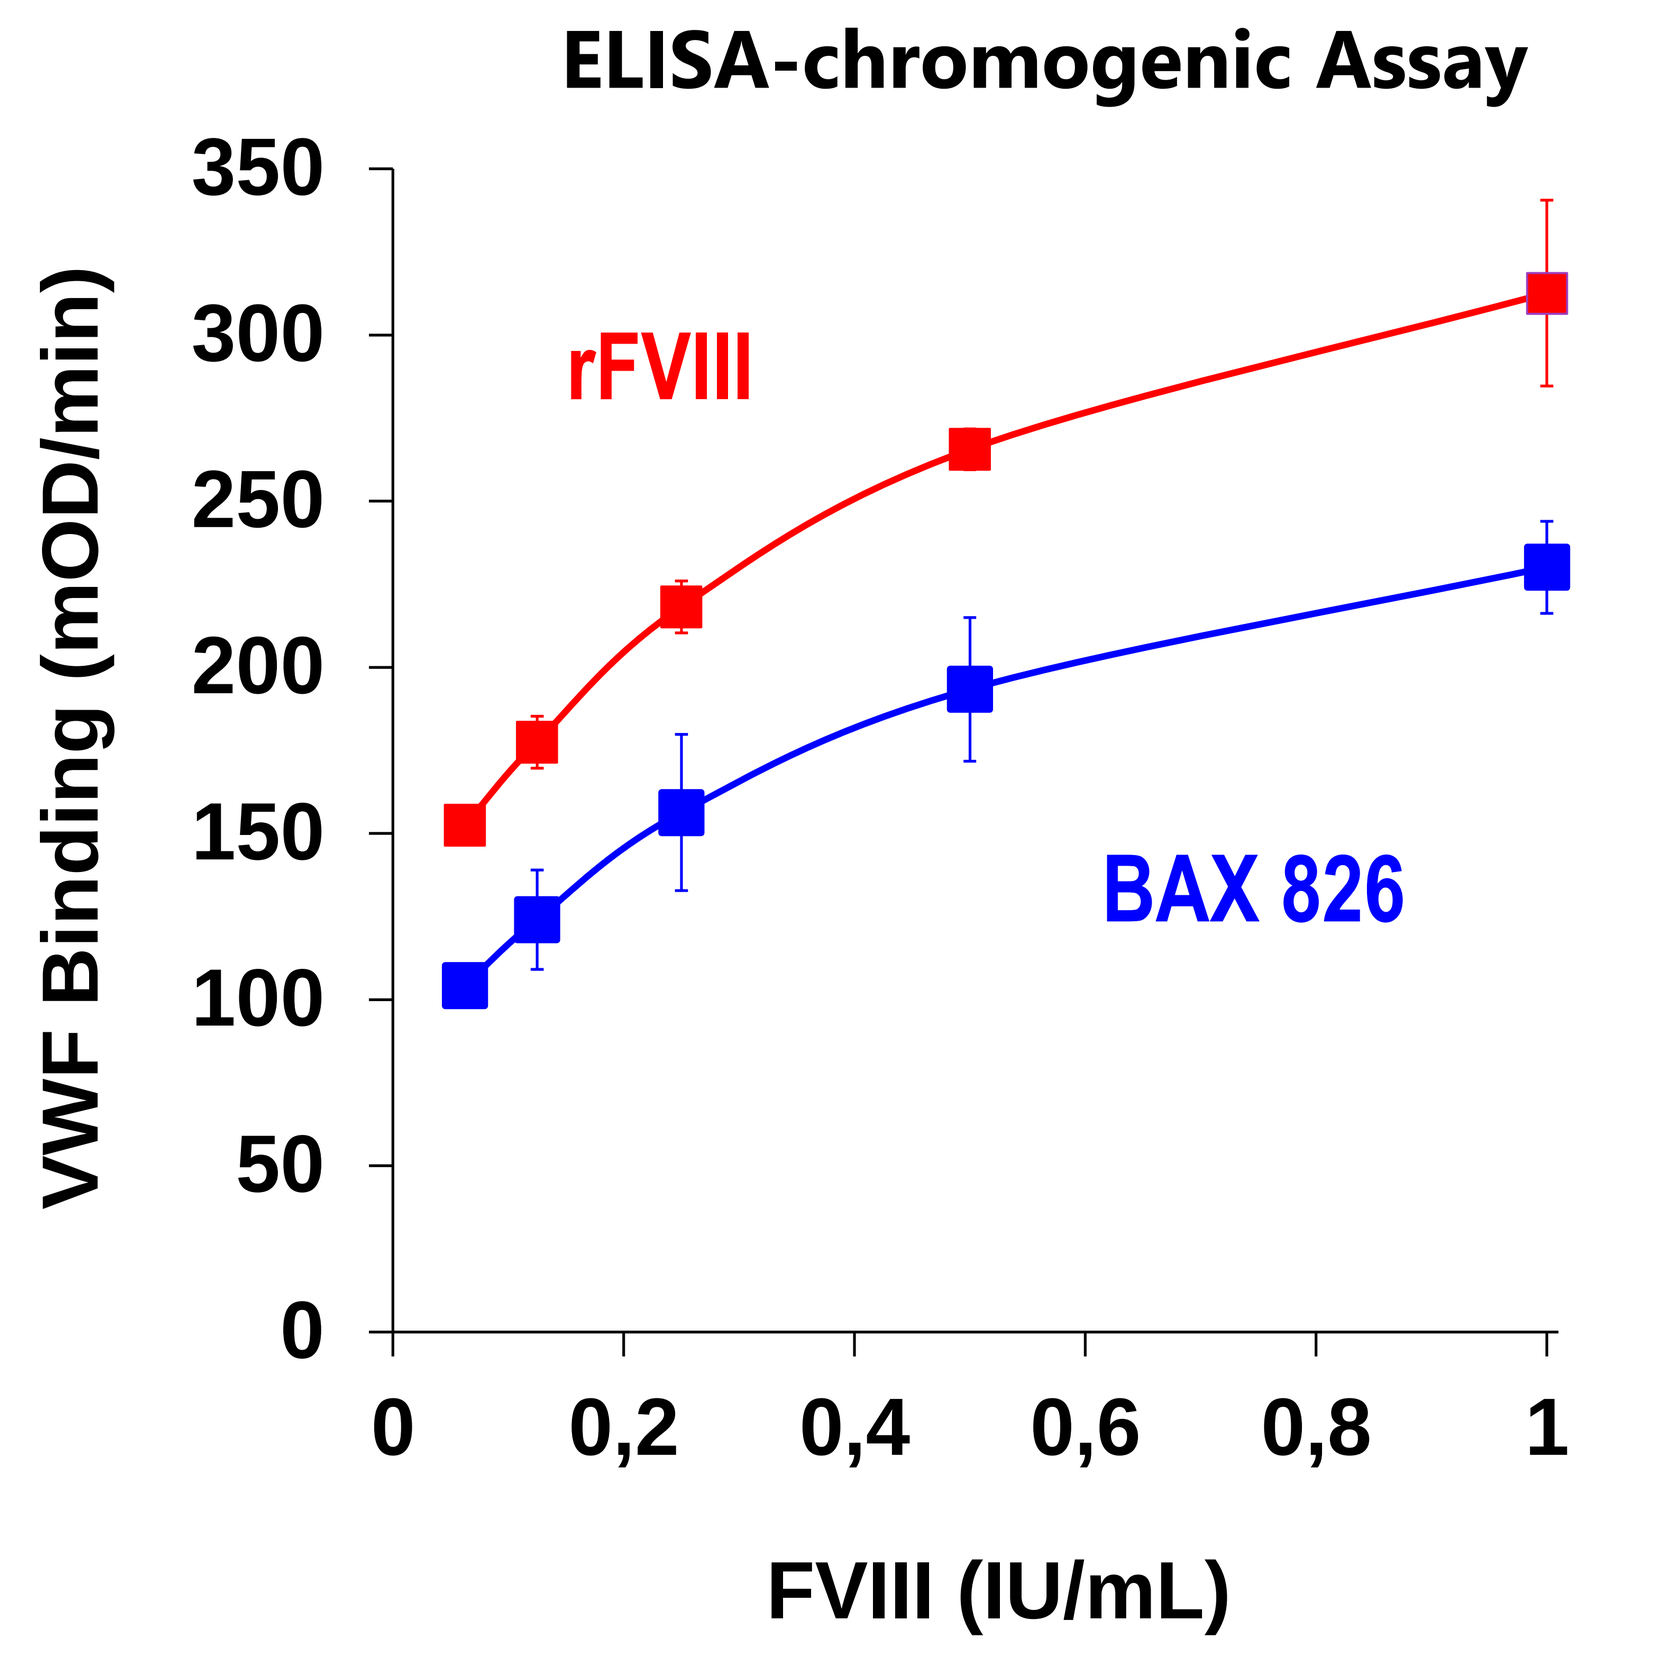
ELISA-chromogenic Assay
VWF Binding (mOD/min)
350
300
250
200
150
100
50
0
rFVIII
BAX 826
0
0,2
0,4
0,6
0,8
1
FVIII (IU/mL)

Figure 4: Results for three independent measurements. Error bars show standard deviation.

- FVIII-VWF complex formation was studied using Biacore and ELISA-combined chromogenic assay.
- In the Biacore assay, the overall binding response of BAX 826 to VWF was lower than to rFVIII.
- Data from the ELISA-based method were in agreement, confirming reduced BAX 826-VWF interaction.
- Similar to FVIII-LRP1, the binding affinity of BAX 826 and rFVIII to VWF was similar, with K_D values ranging from 0.24 to 0.39 nM for BAX 826 (n = 10) and from 0.26 to 0.37 nM for rFVIII (n = 4).
- These results demonstrate that modifying rFVIII with PSA also affects binding to FVIII´s chaperone protein VWF.

RESULTS – Pharmacokinetics in FVIIIxVWF Double Knockout Mice

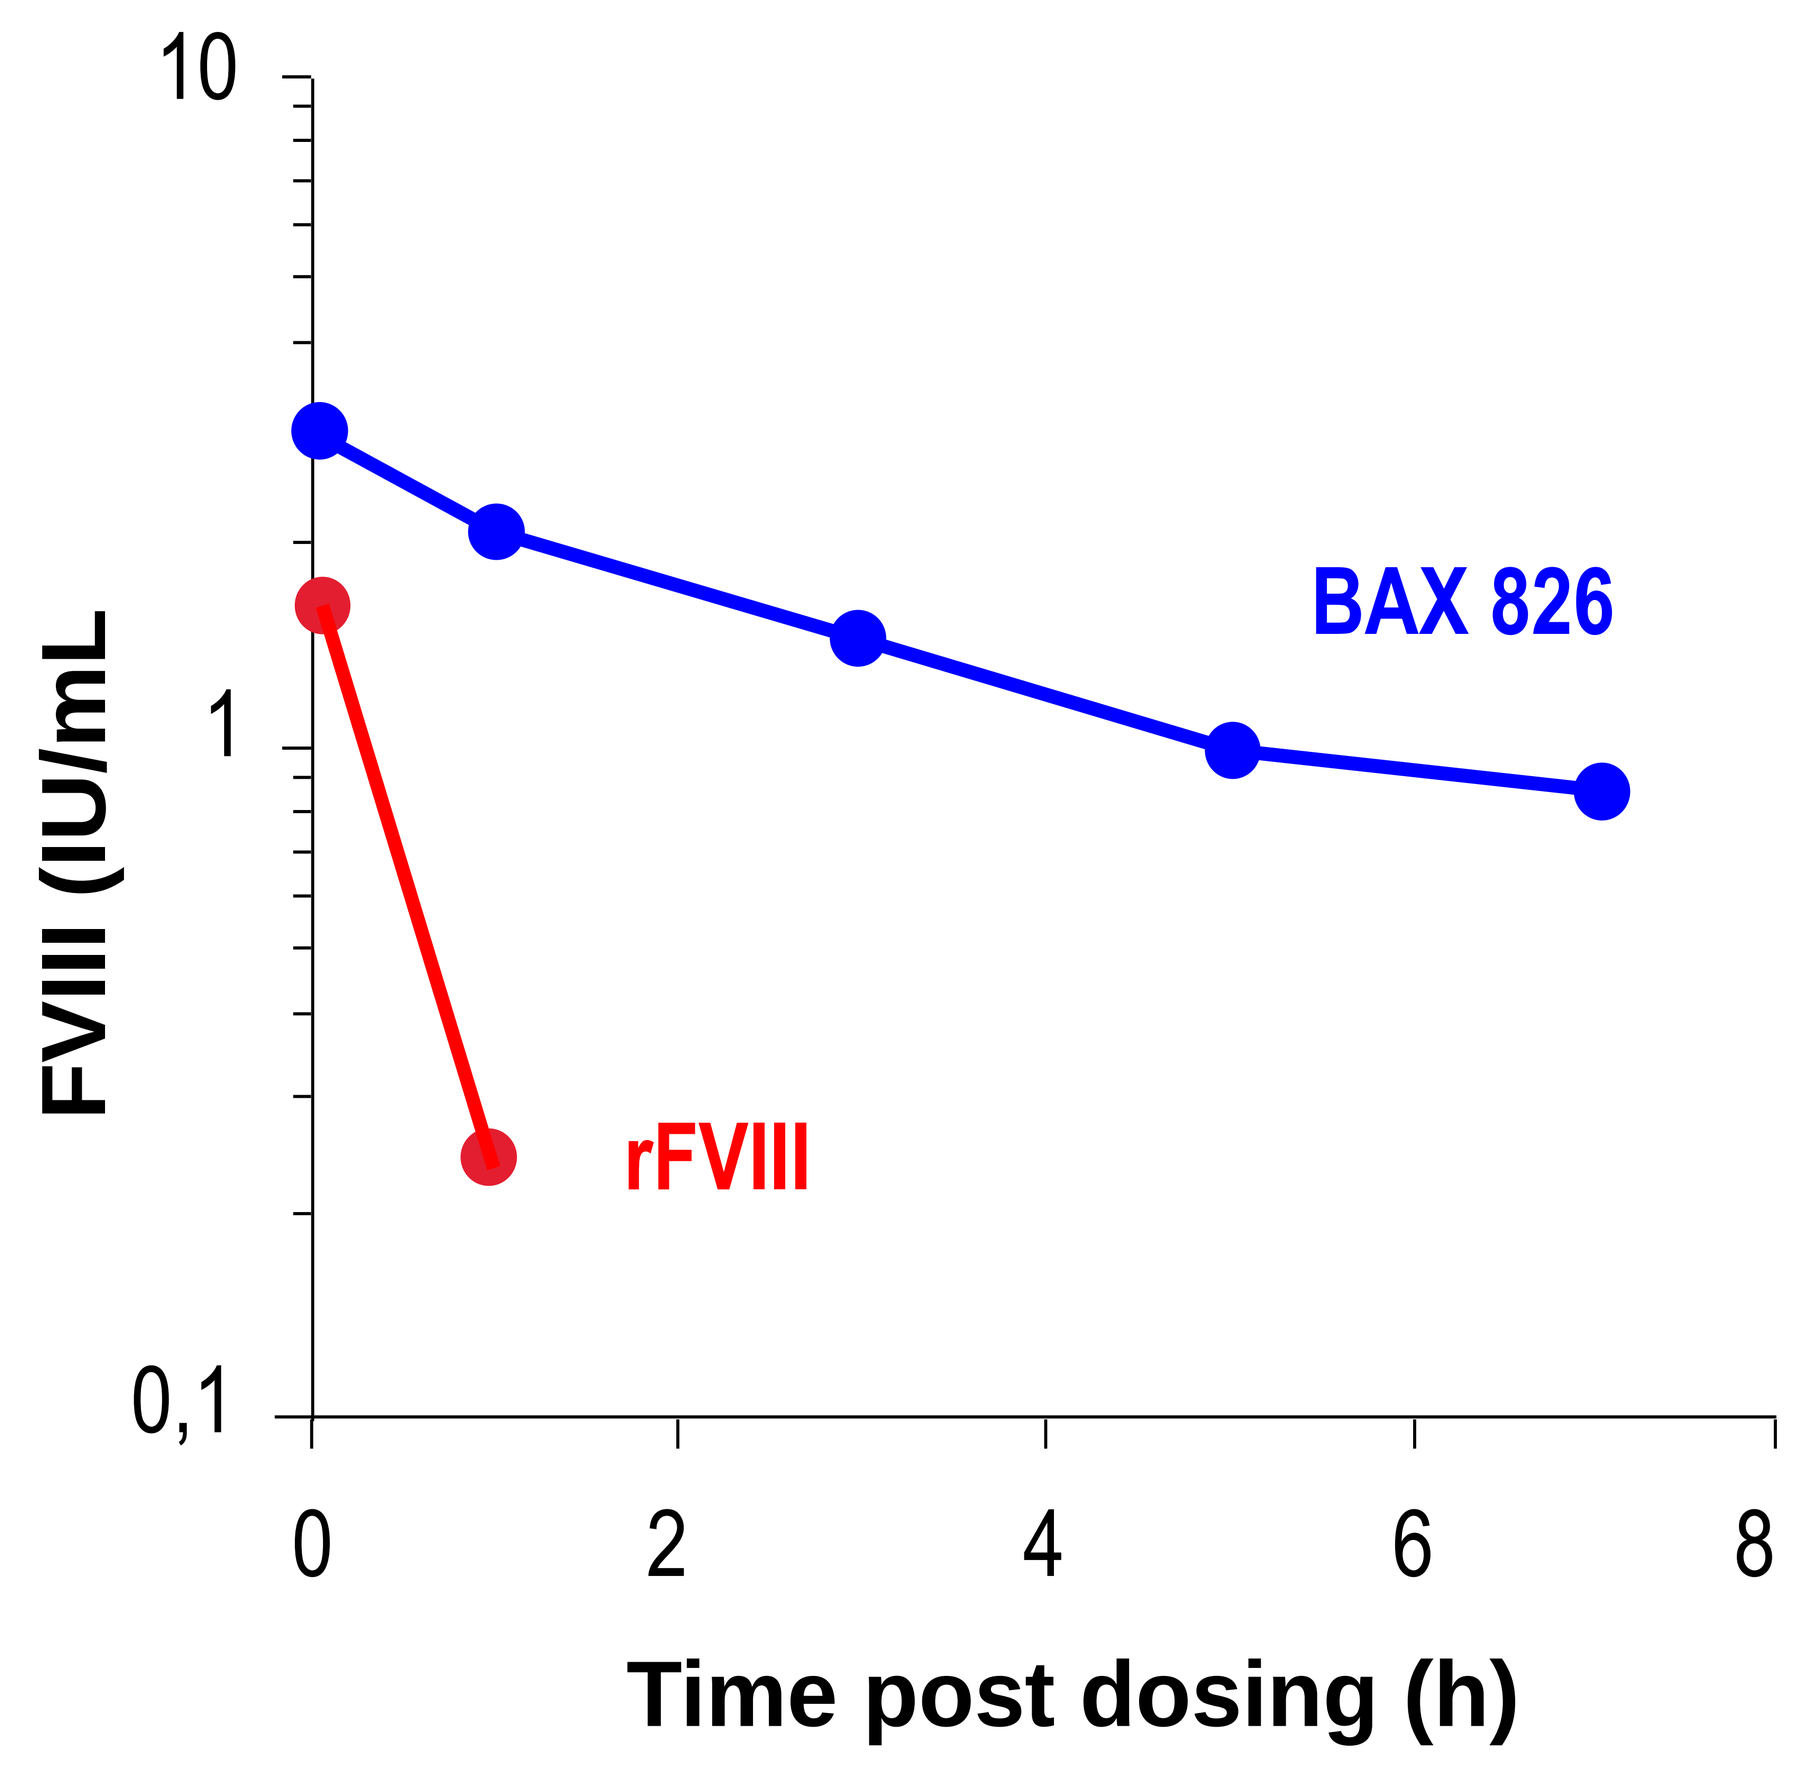
FVIII (IU/mL
10
1
0,1
BAX 826
rFVIII
0
2
4
6
8
Time post dosing (h)

PK parameter	rFVIII	BAX 826
Max. concentration (IU/mL)	**1.6**	**3.4**
in vivo recovery (%)	**39.6**	**71.3**
Terminal half-life (h)	**0.3***	**5.5**
Mean residence time (h)	**na**	**7.7**

*Assuming that the terminal phase for rFVIII is adequately represented by concentrations observed at 5 min and 1 h only.

- Due to the absence of VWF, rFVIII was rapidly cleared from the circulation and undetectable 3 h after injection. The estimated half-life was 0.3 h.
 In contrast, the terminal half-life of BAX 826 was prolonged to 5.5 h in this mouse model, suggesting a lower clearance of BAX 826 in the absence of the chaperone VWF.

CONCLUSION

- Two independent *in vitro* methods demonstrated that modifying rFVIII with PSA markedly decreased its interaction with LRP1, an important clearance receptor for FVIII.
- Binding of BAX 826 to FVIII´s chaperone protein VWF was also reduced, which could render BAX 826 independent of clearance of the VWF-FVIII complex, likely escaping the half life limitation determined by VWF.
- BAX 826 showed a substantially longer circulation time in a selective FVIIIxVWF double knockout mouse model system.
- In conclusion, these results qualify BAX 826 as an attractive new extended half-life FVIII candidate with an alternative mechanism for half-life prolongation. Extensive clinical investigation of BAX 826 is required to further address these findings.

DISCLOSURES

*All authors are employees of Baxalta (Baxalta Innovations GmbH), now part of Shire.

WFH World Congress; July 24–28, 2016; Orlando, USA

PosterSessionOnline.com
Poster Presented at:
WFH 2016 World Congress Orlando, USA July 24-28
DOI: 10.3252/pso.eu.WFH2016.2016
Novel therapeutic agents
Michael Dockal
89--PP-M
WFH2016