POSTER #190

FVII Annual Retreats Help Guide Patients & Families to a Better Understanding of Living With a Rare Bleeding Disorder

by Janet H Brewer M.Ed and Barb Forss

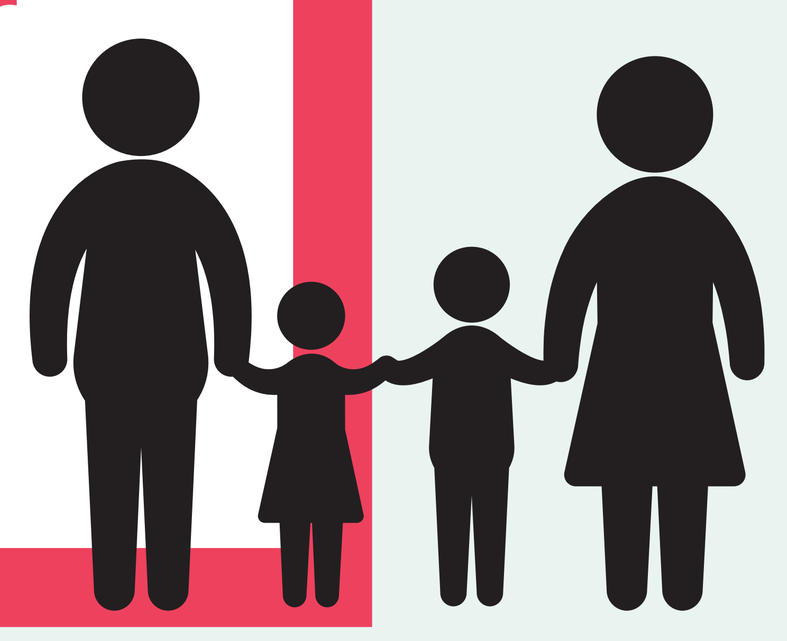

"We share information about retreats in newsletters, websites and social media."

The group has informally named themselves the:

SUPER SEVENS

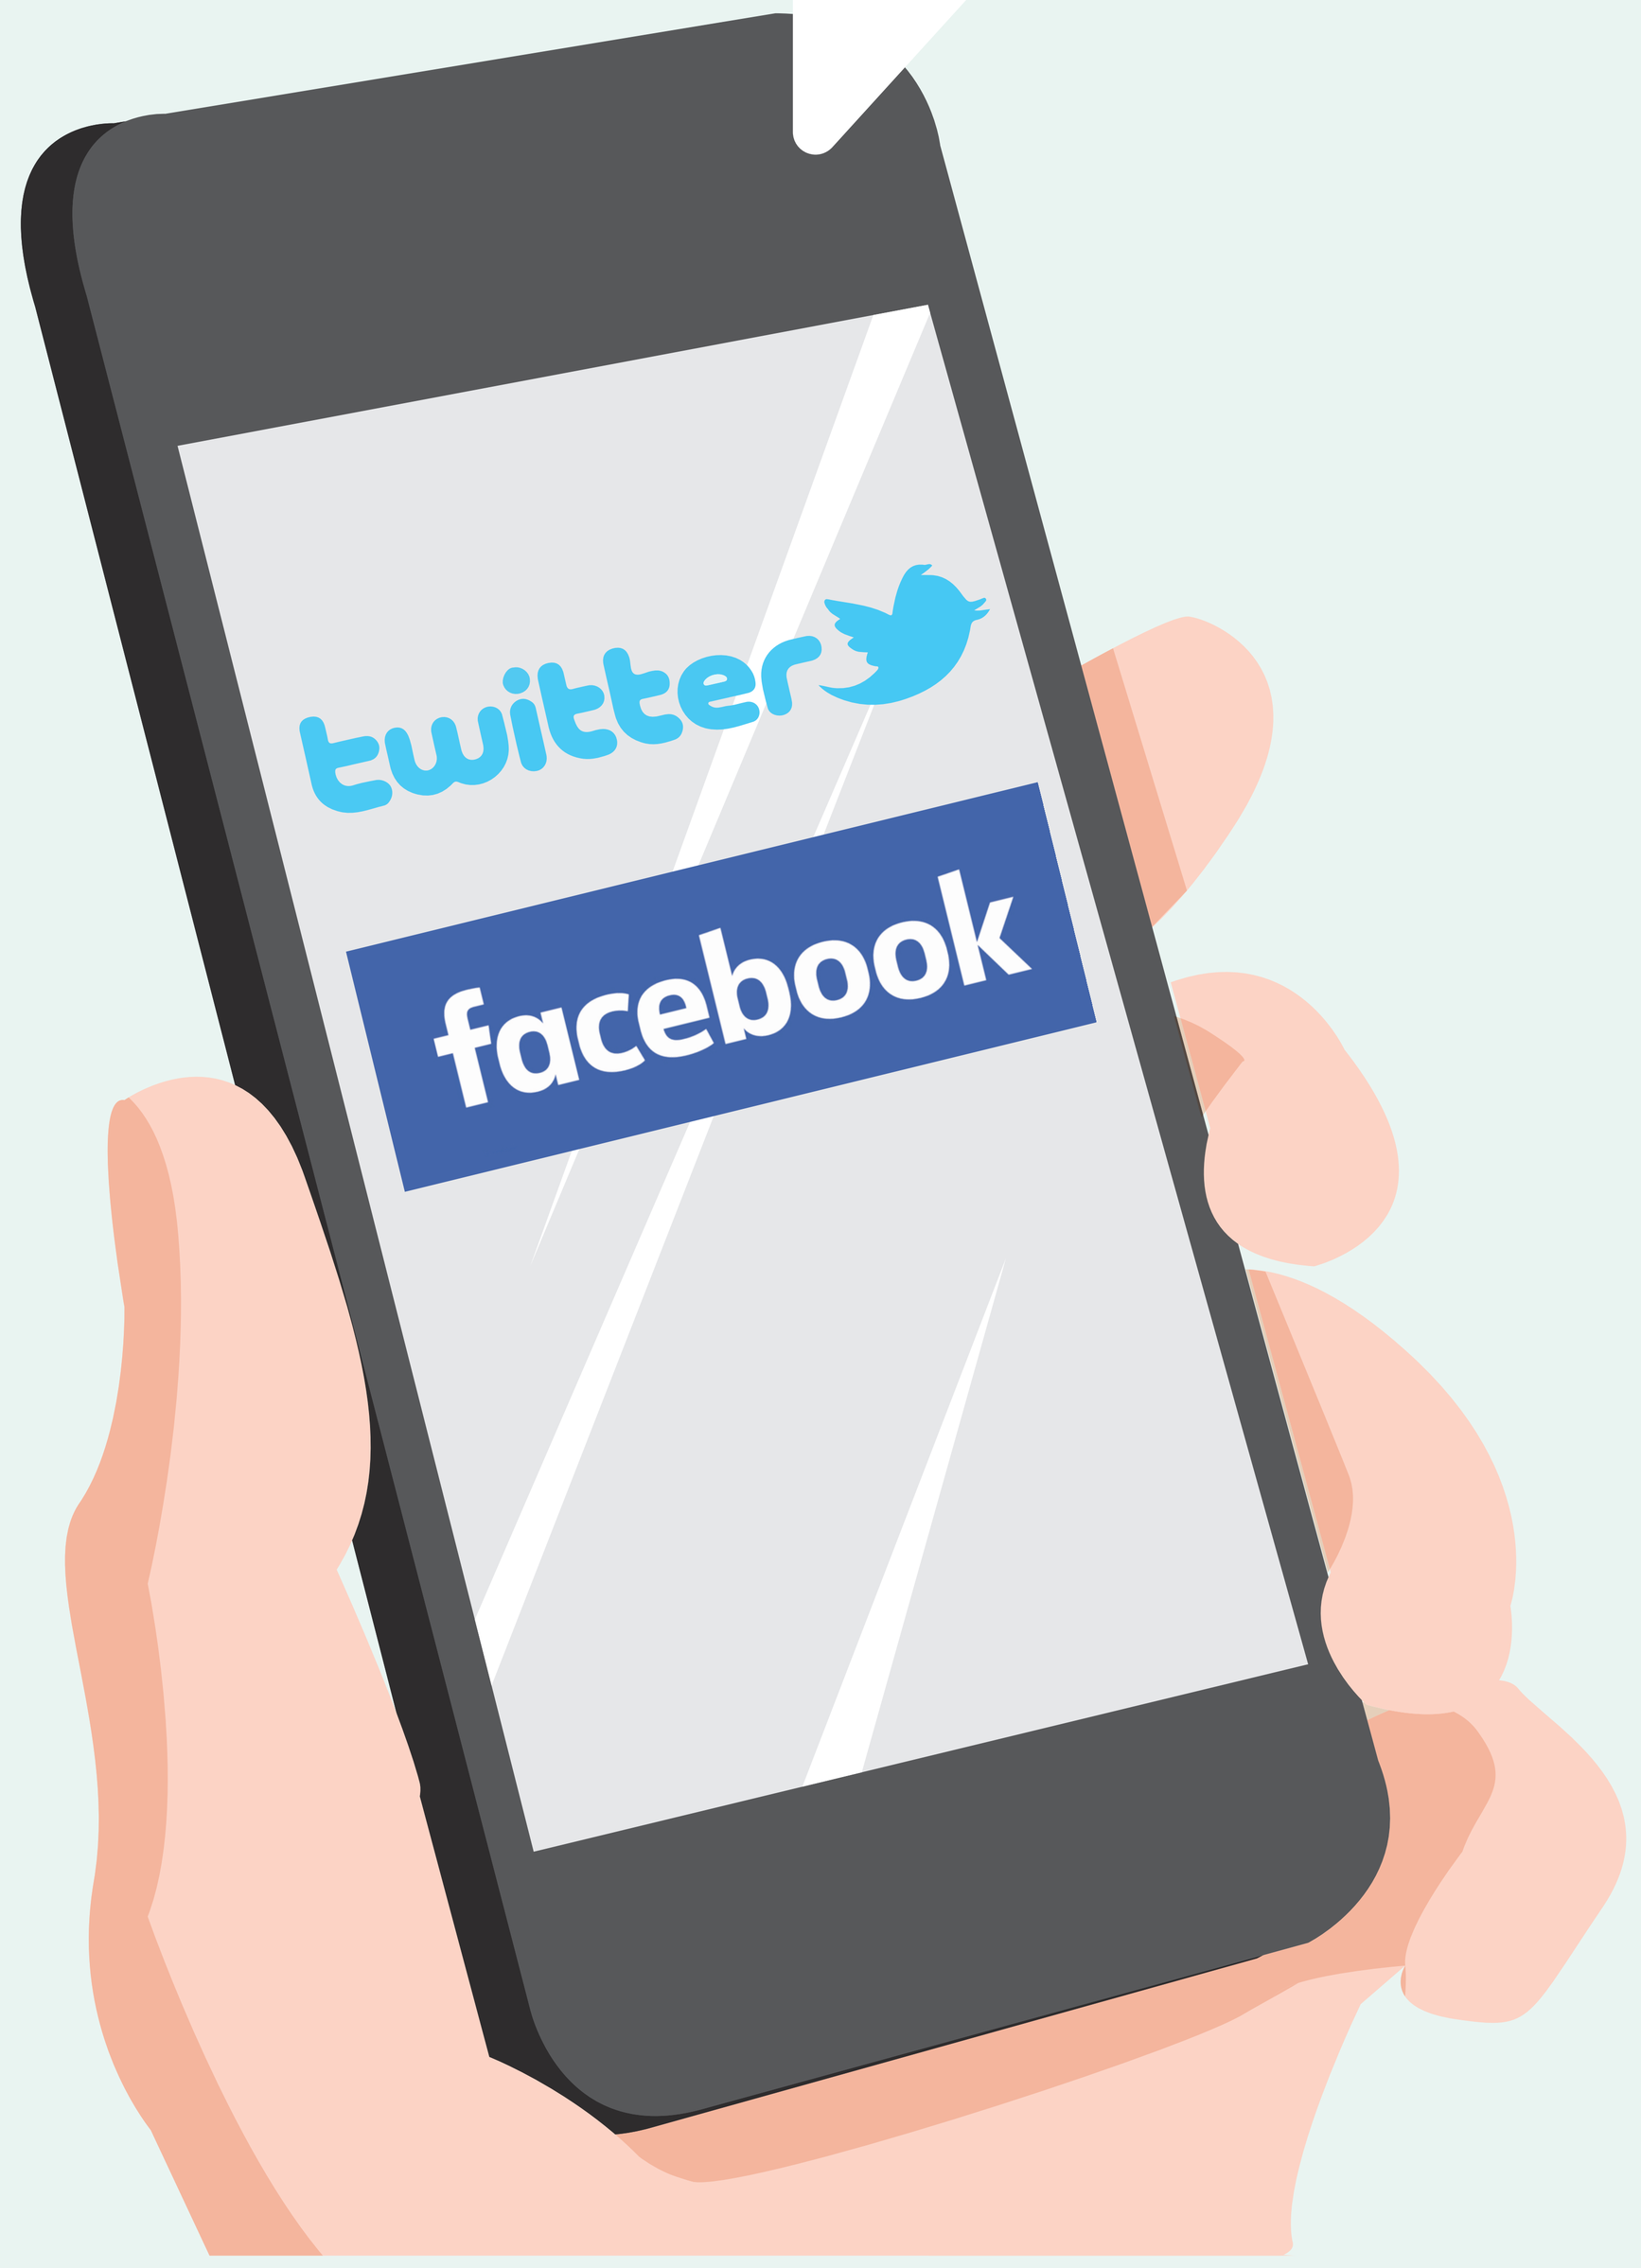
twitter
facebook

INTRODUCTION & OBJECTIVES

Categorized as a Rare Bleeding Disorder (RBD), fVIId is the third most common factor deficiency, yet very few educational opportunities exist for these patients and their families to meet and learn from providers and each other. The need for a program that provides support for patients with fVIId is clear.

MATERIALS & METHODS

From 2012-2015, Comprehensive Health Education Services and the LadyBugs Foundation have collaborated to provide a 3-day program for patients and their families, many of which have several affected members with fVIId.

Physicians, nurse infusion specialists, physical therapists, insurance experts, psychologists, exercise leaders and others have been invited to present sessions about improving the lives of those with fVIId and their families.

Our patient recruitment methods include outreach to all National Hemophilia Chapters, Hemophilia Treatment Centers and Home Health Care Companies, with a request to share information about the retreat in their newsletters, websites and social media.

Our organizations distribute information via physical mailings and social media. Requests are made to HFA and NHF to include information regarding the FVII Retreat in their social media posits, newsletters and mailing lists.

RESULTS

Since 2012, attendance at the FVII Retreat has increased by 128%. Surveyed attendees indicate that the three most important parts of the program are social networking, education on bleeding tendencies/patterns/treatment, and self-infusion classes.

Their most common challenges are lack of communication, awareness and understanding of fVIId.

Overall program satisfaction rating is: **95%**

Overall session satisfaction rating is: **94%**

In four sessions, we served:

78 Unique Patients

176 Patients*

482 Individuals

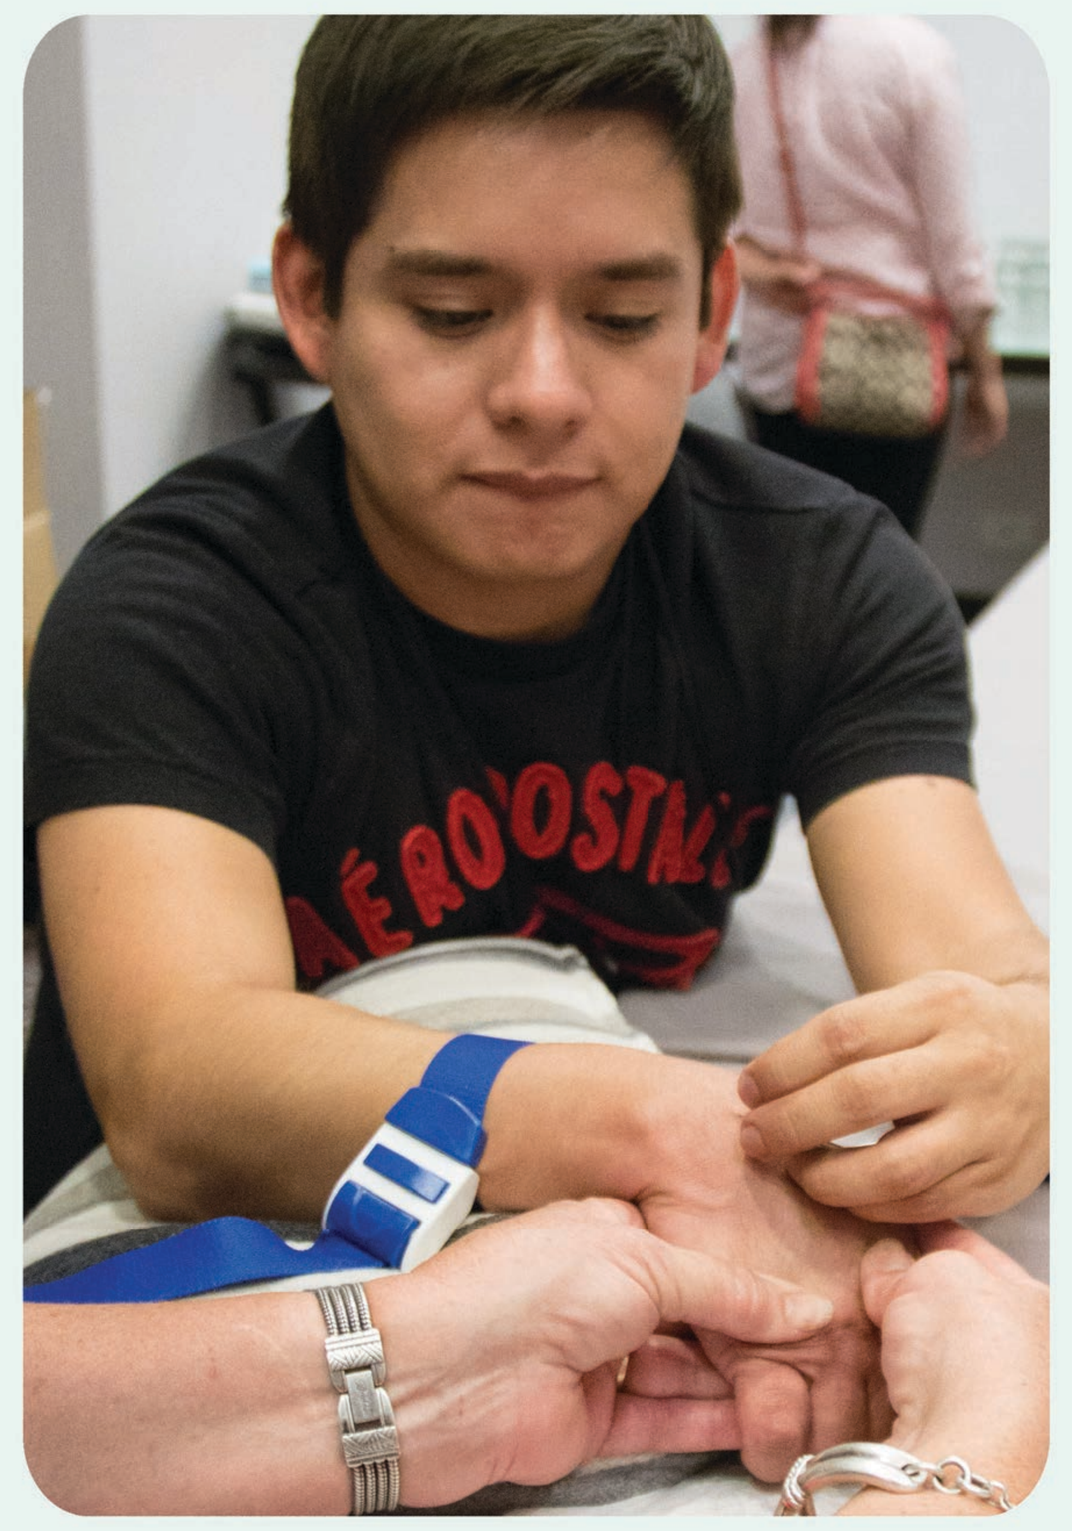

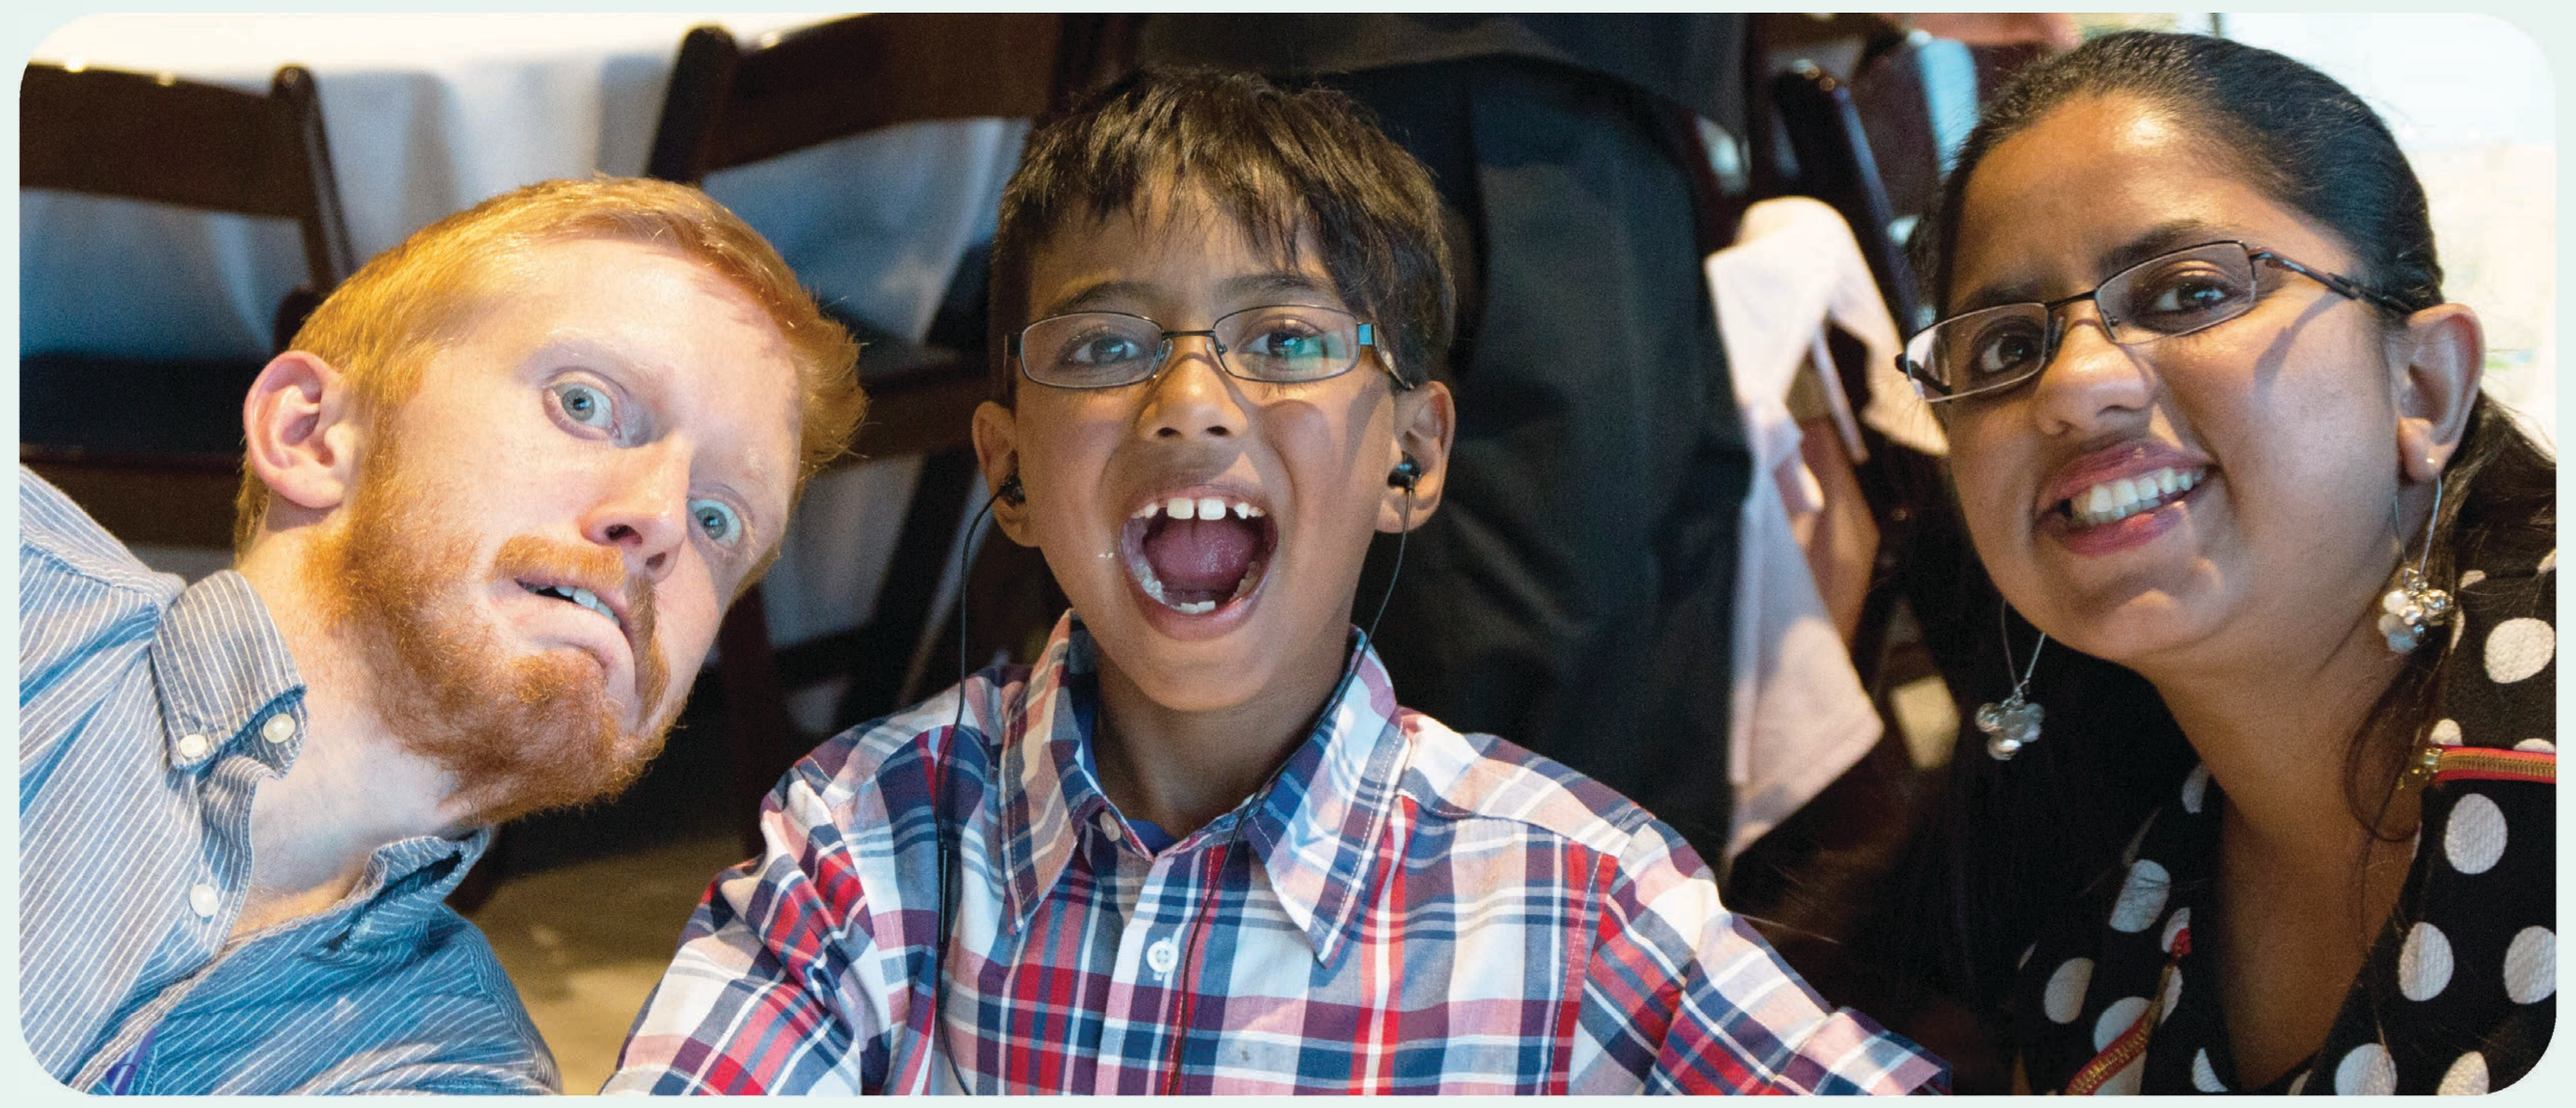
AÉROPOSTALE

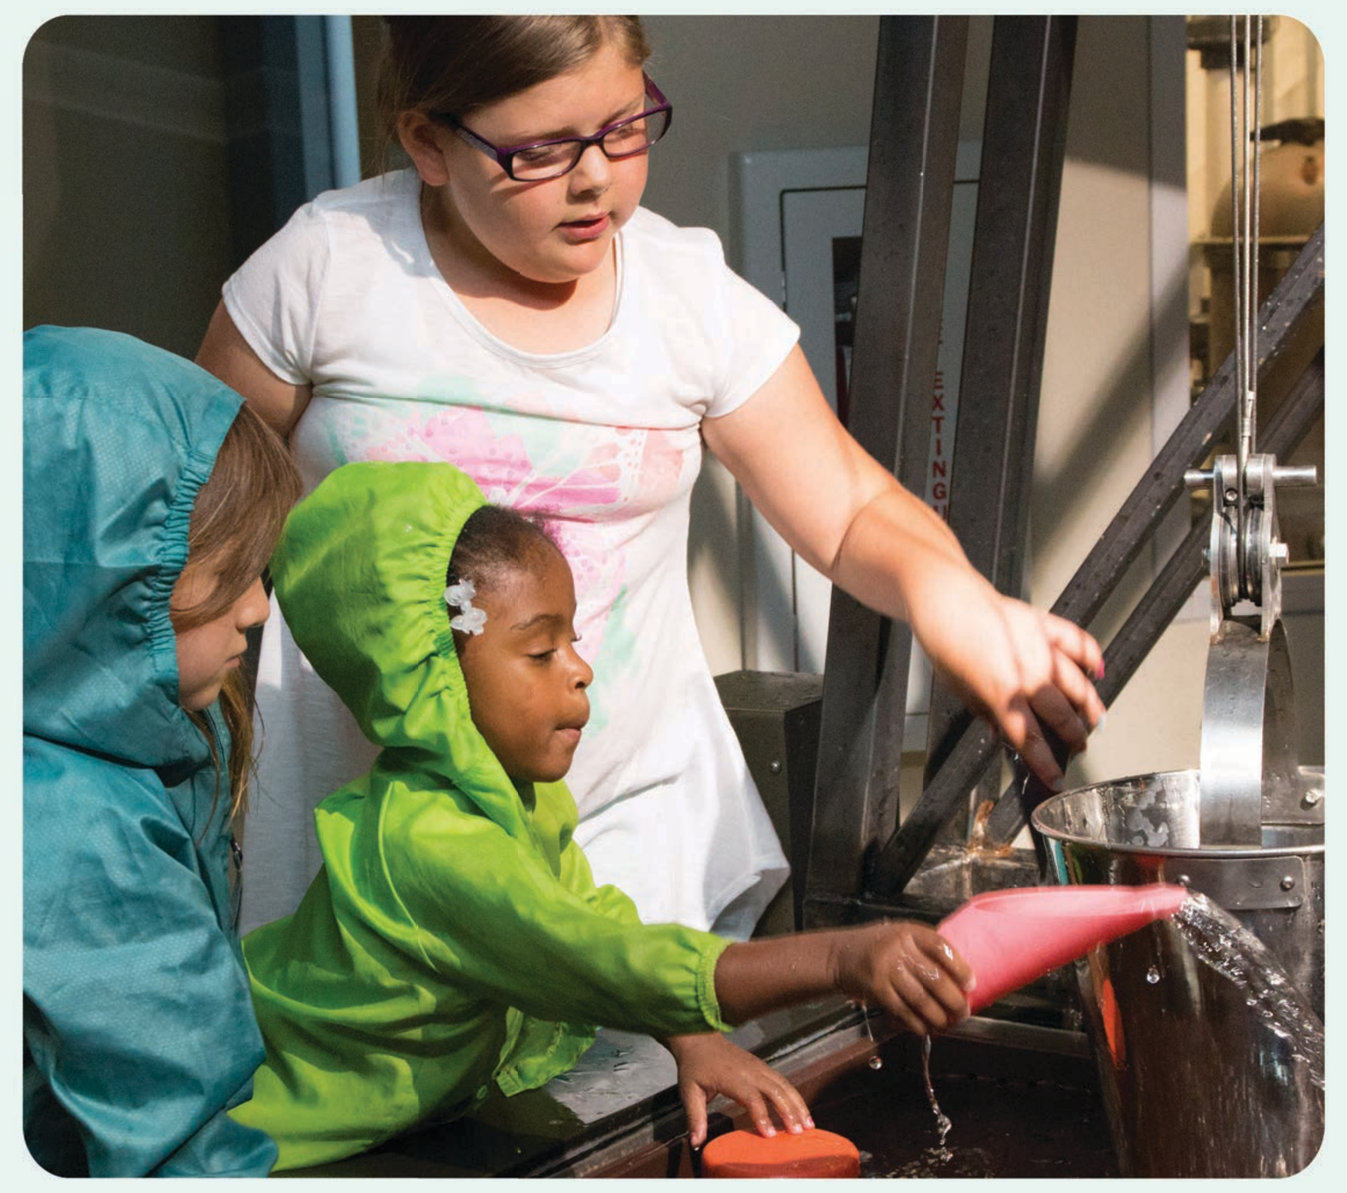

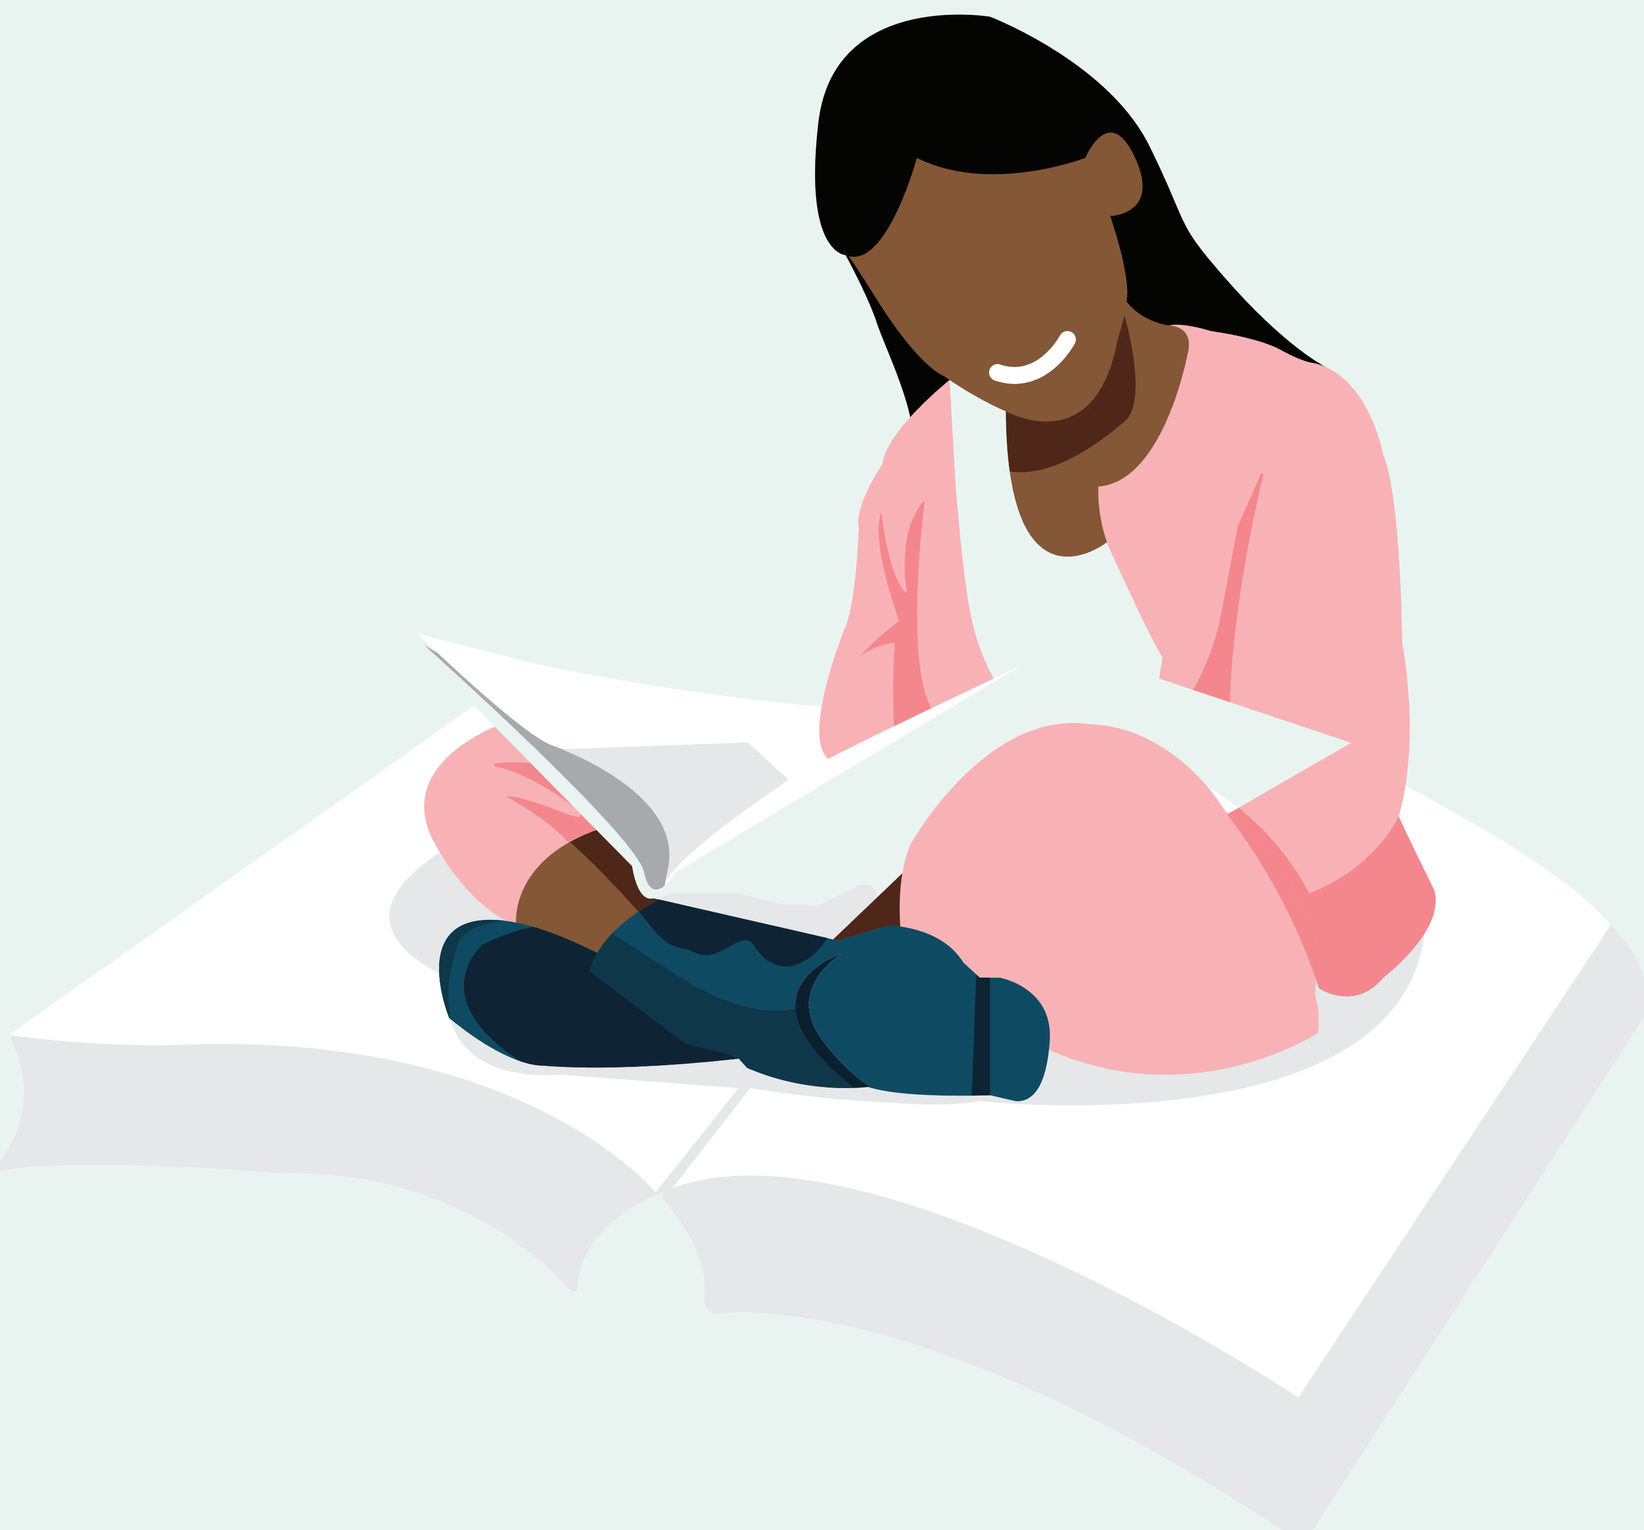

CONCLUSIONS

Annual retreats for patients and families affected by fVIId are a vital necessity in identifying and helping to meet their needs.

Attendees become better educated and more empowered after attending this retreat. They are able to establish a peer network that diminishes some of the isolation they feel to improve overall quality of life.

One participant commented:

I feel I have a life preserver and I am not under water anymore. This is the first time since my daughter has been diagnosed that I can be optimistic. Thank you!

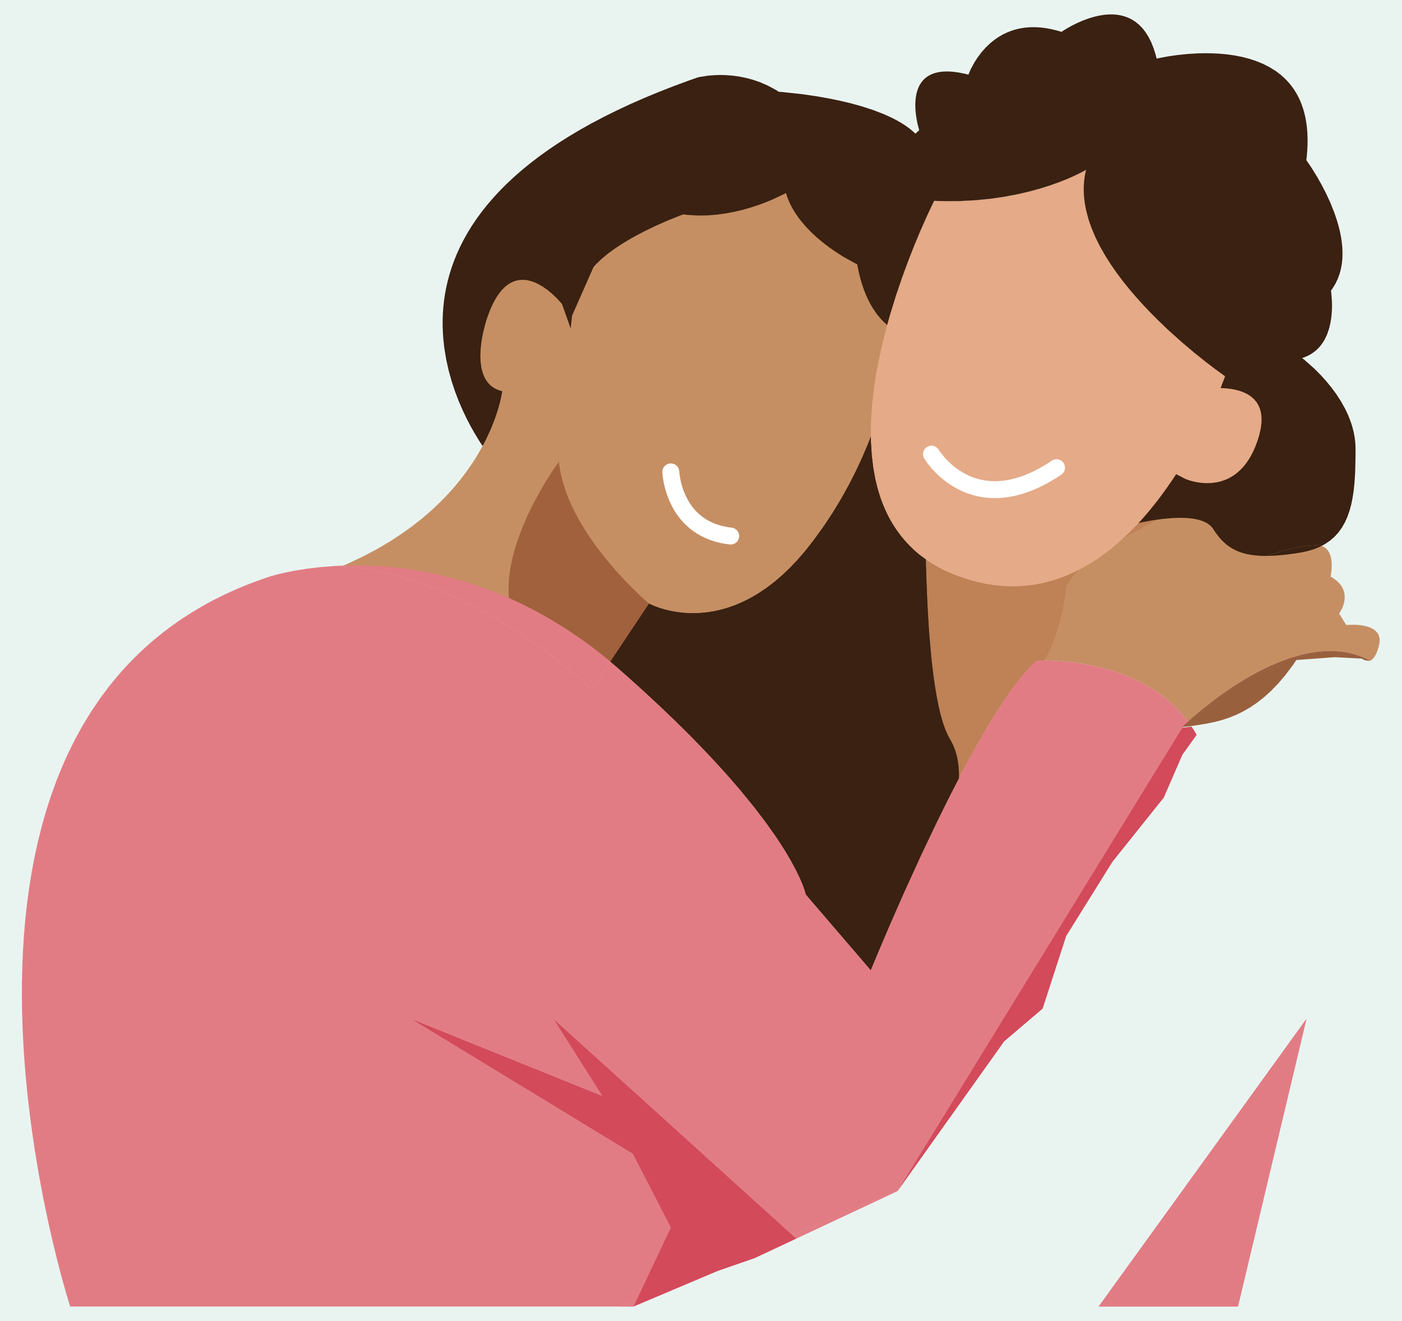

Conflicts of interest and all outside sources of funding

The author and owners of Comprehensive Health Education Services receive educational grants sponsored by Novo Nordisk, Baxalta, Grifols, Kedrion for their programming. The FVII Retreat is supported by an educational grant from Novo Nordisk.

Janet Brewer and Barb Forss have both been on consumer advisory boards for Novo Nordisk. Barb has also been featured in promotional materials developed by Novo Nordisk.

*Includes multiple attendances

PosterSessionOnline.com
Poster Presented at:
WFH 2016 World Congress
Orlando, USA, July 24-28
DOI: 10.3252/pso.eu.WFH2016.2016
Quality of Life
Janet Brewer
190--PP-M
WFH2016