POSTER #187

Inhibitor Family Camp Provides a Safe Haven for Children & Families to Have a Camp Experience

by Janet H Brewer M.Ed and Eric G. Lowe

Introduction and Objectives

Inhibitor Family Camp (IFC) was first established as parents and children in the inhibitor community expressed the need to interact in a more intimate environment. Camp is a place where children (and adults) build social competence and increased self-identity by participating in adventuresome outdoor activities and challenging themselves to try something new.

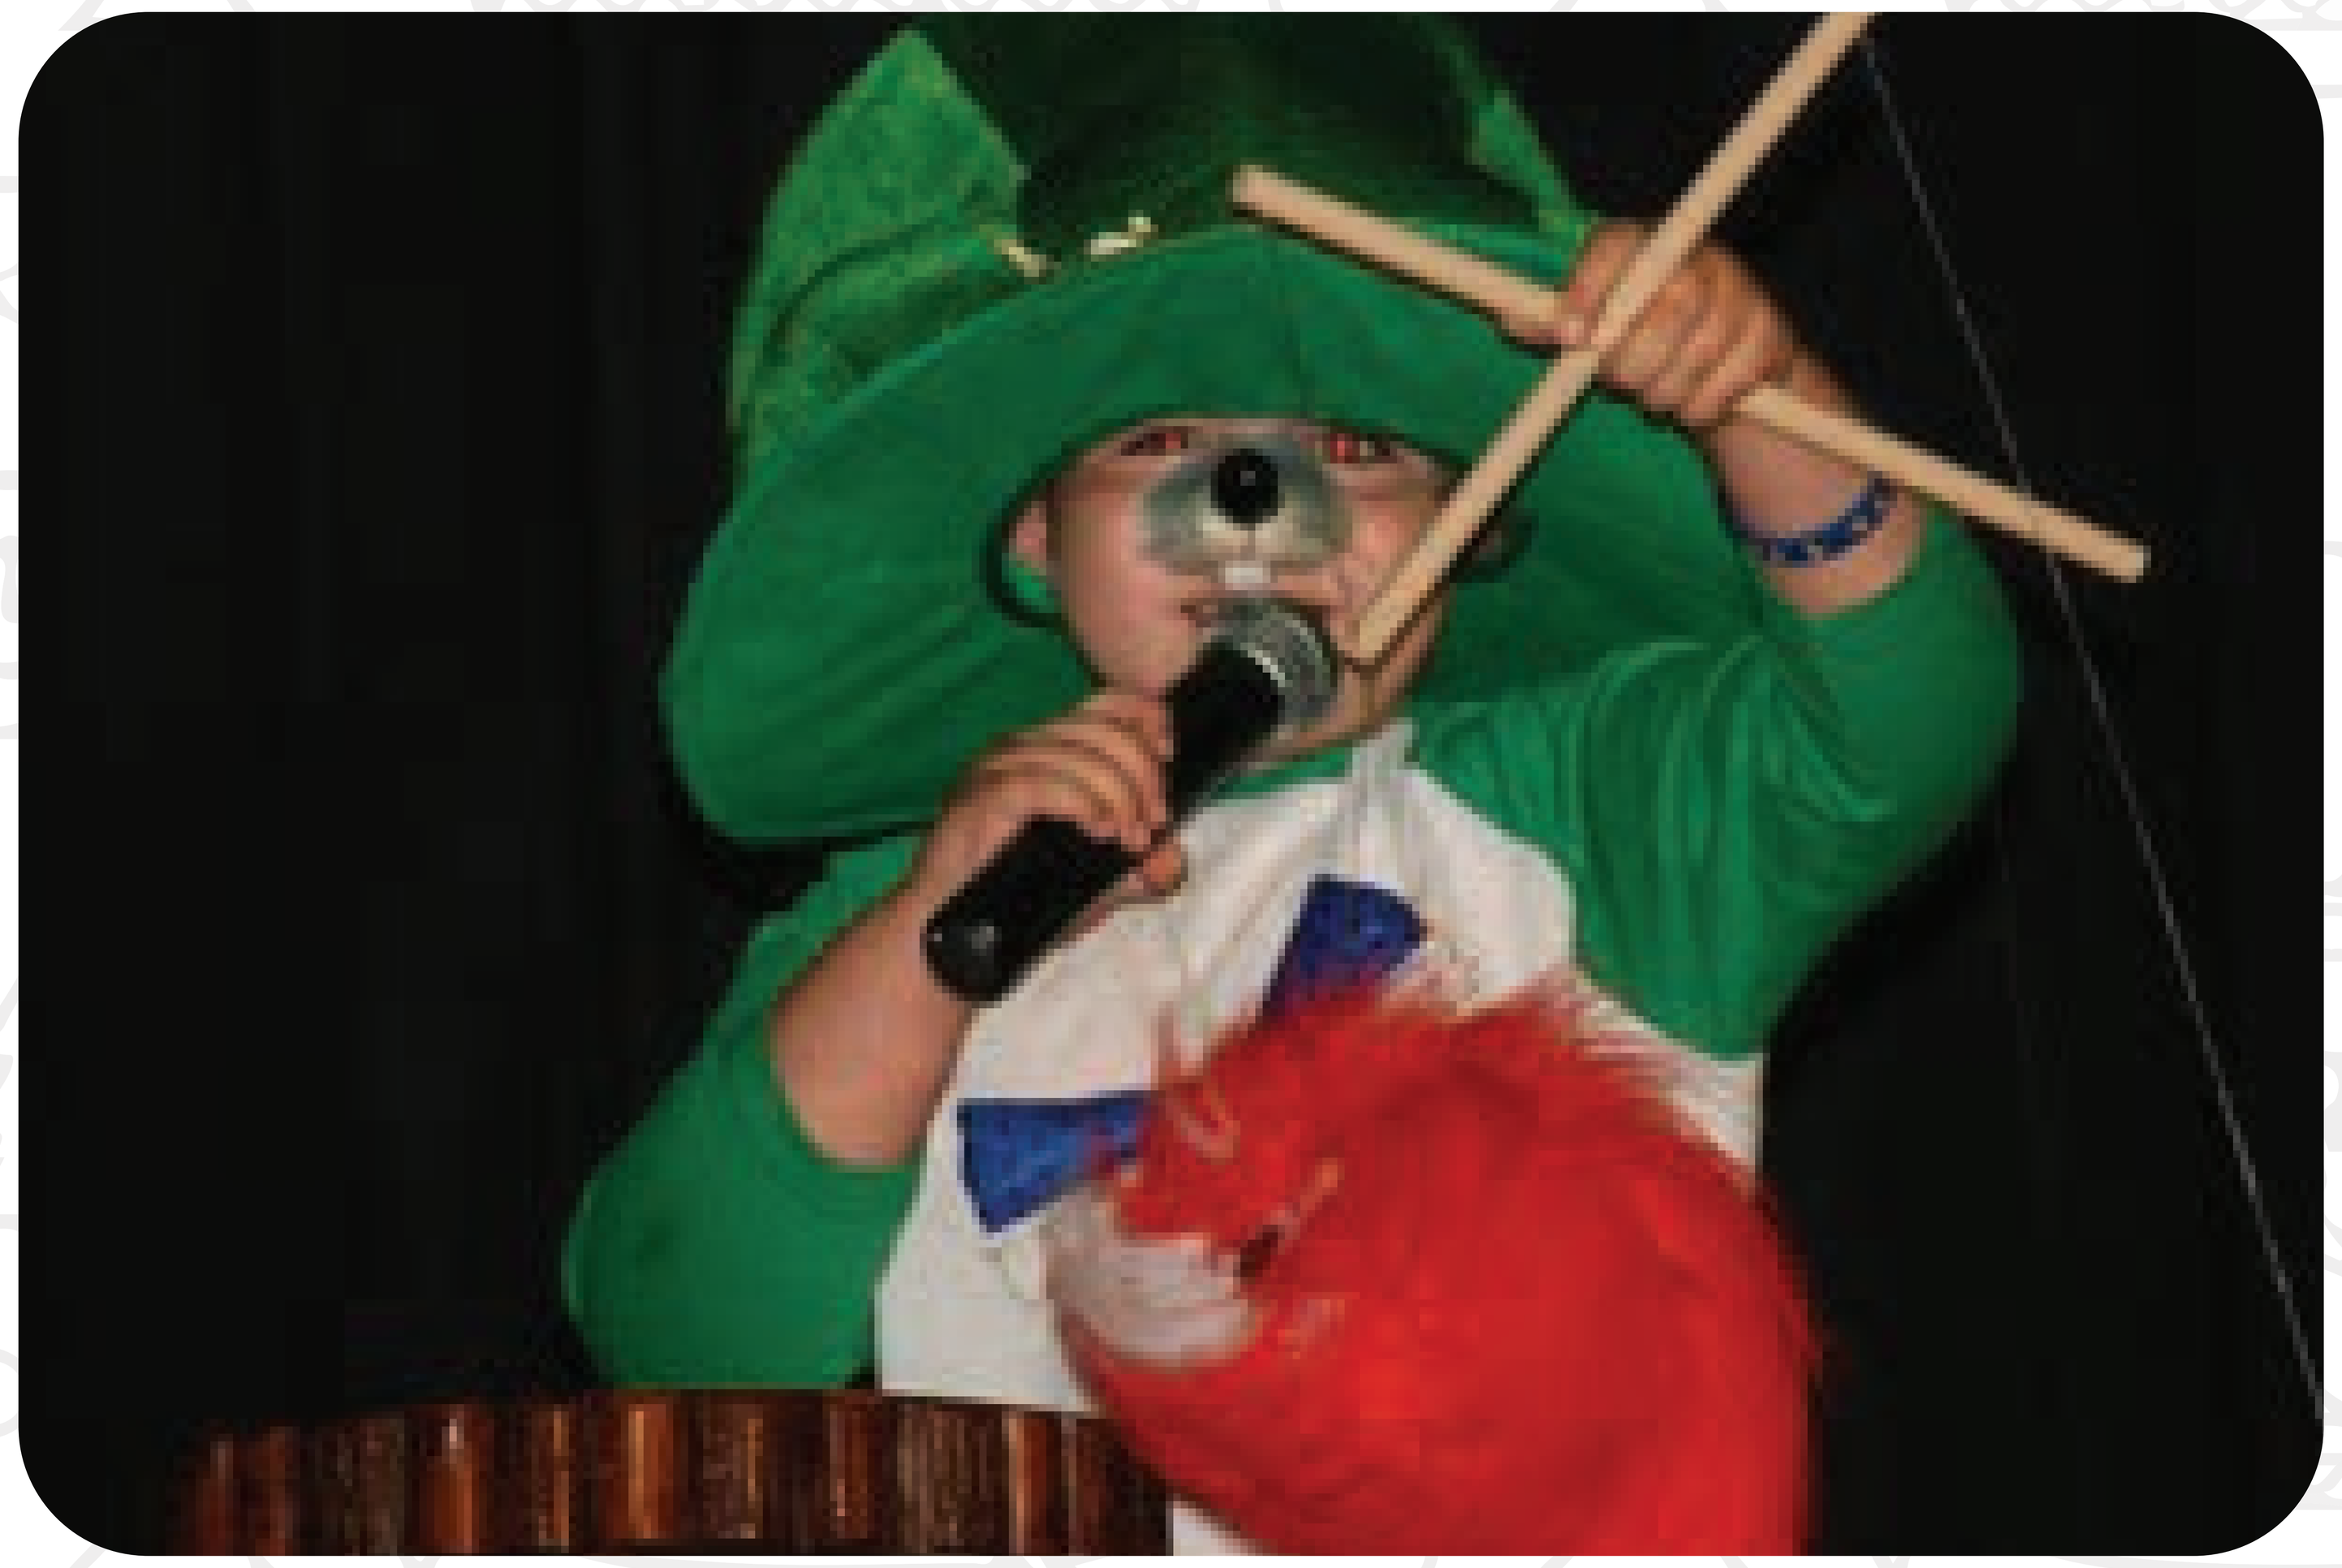

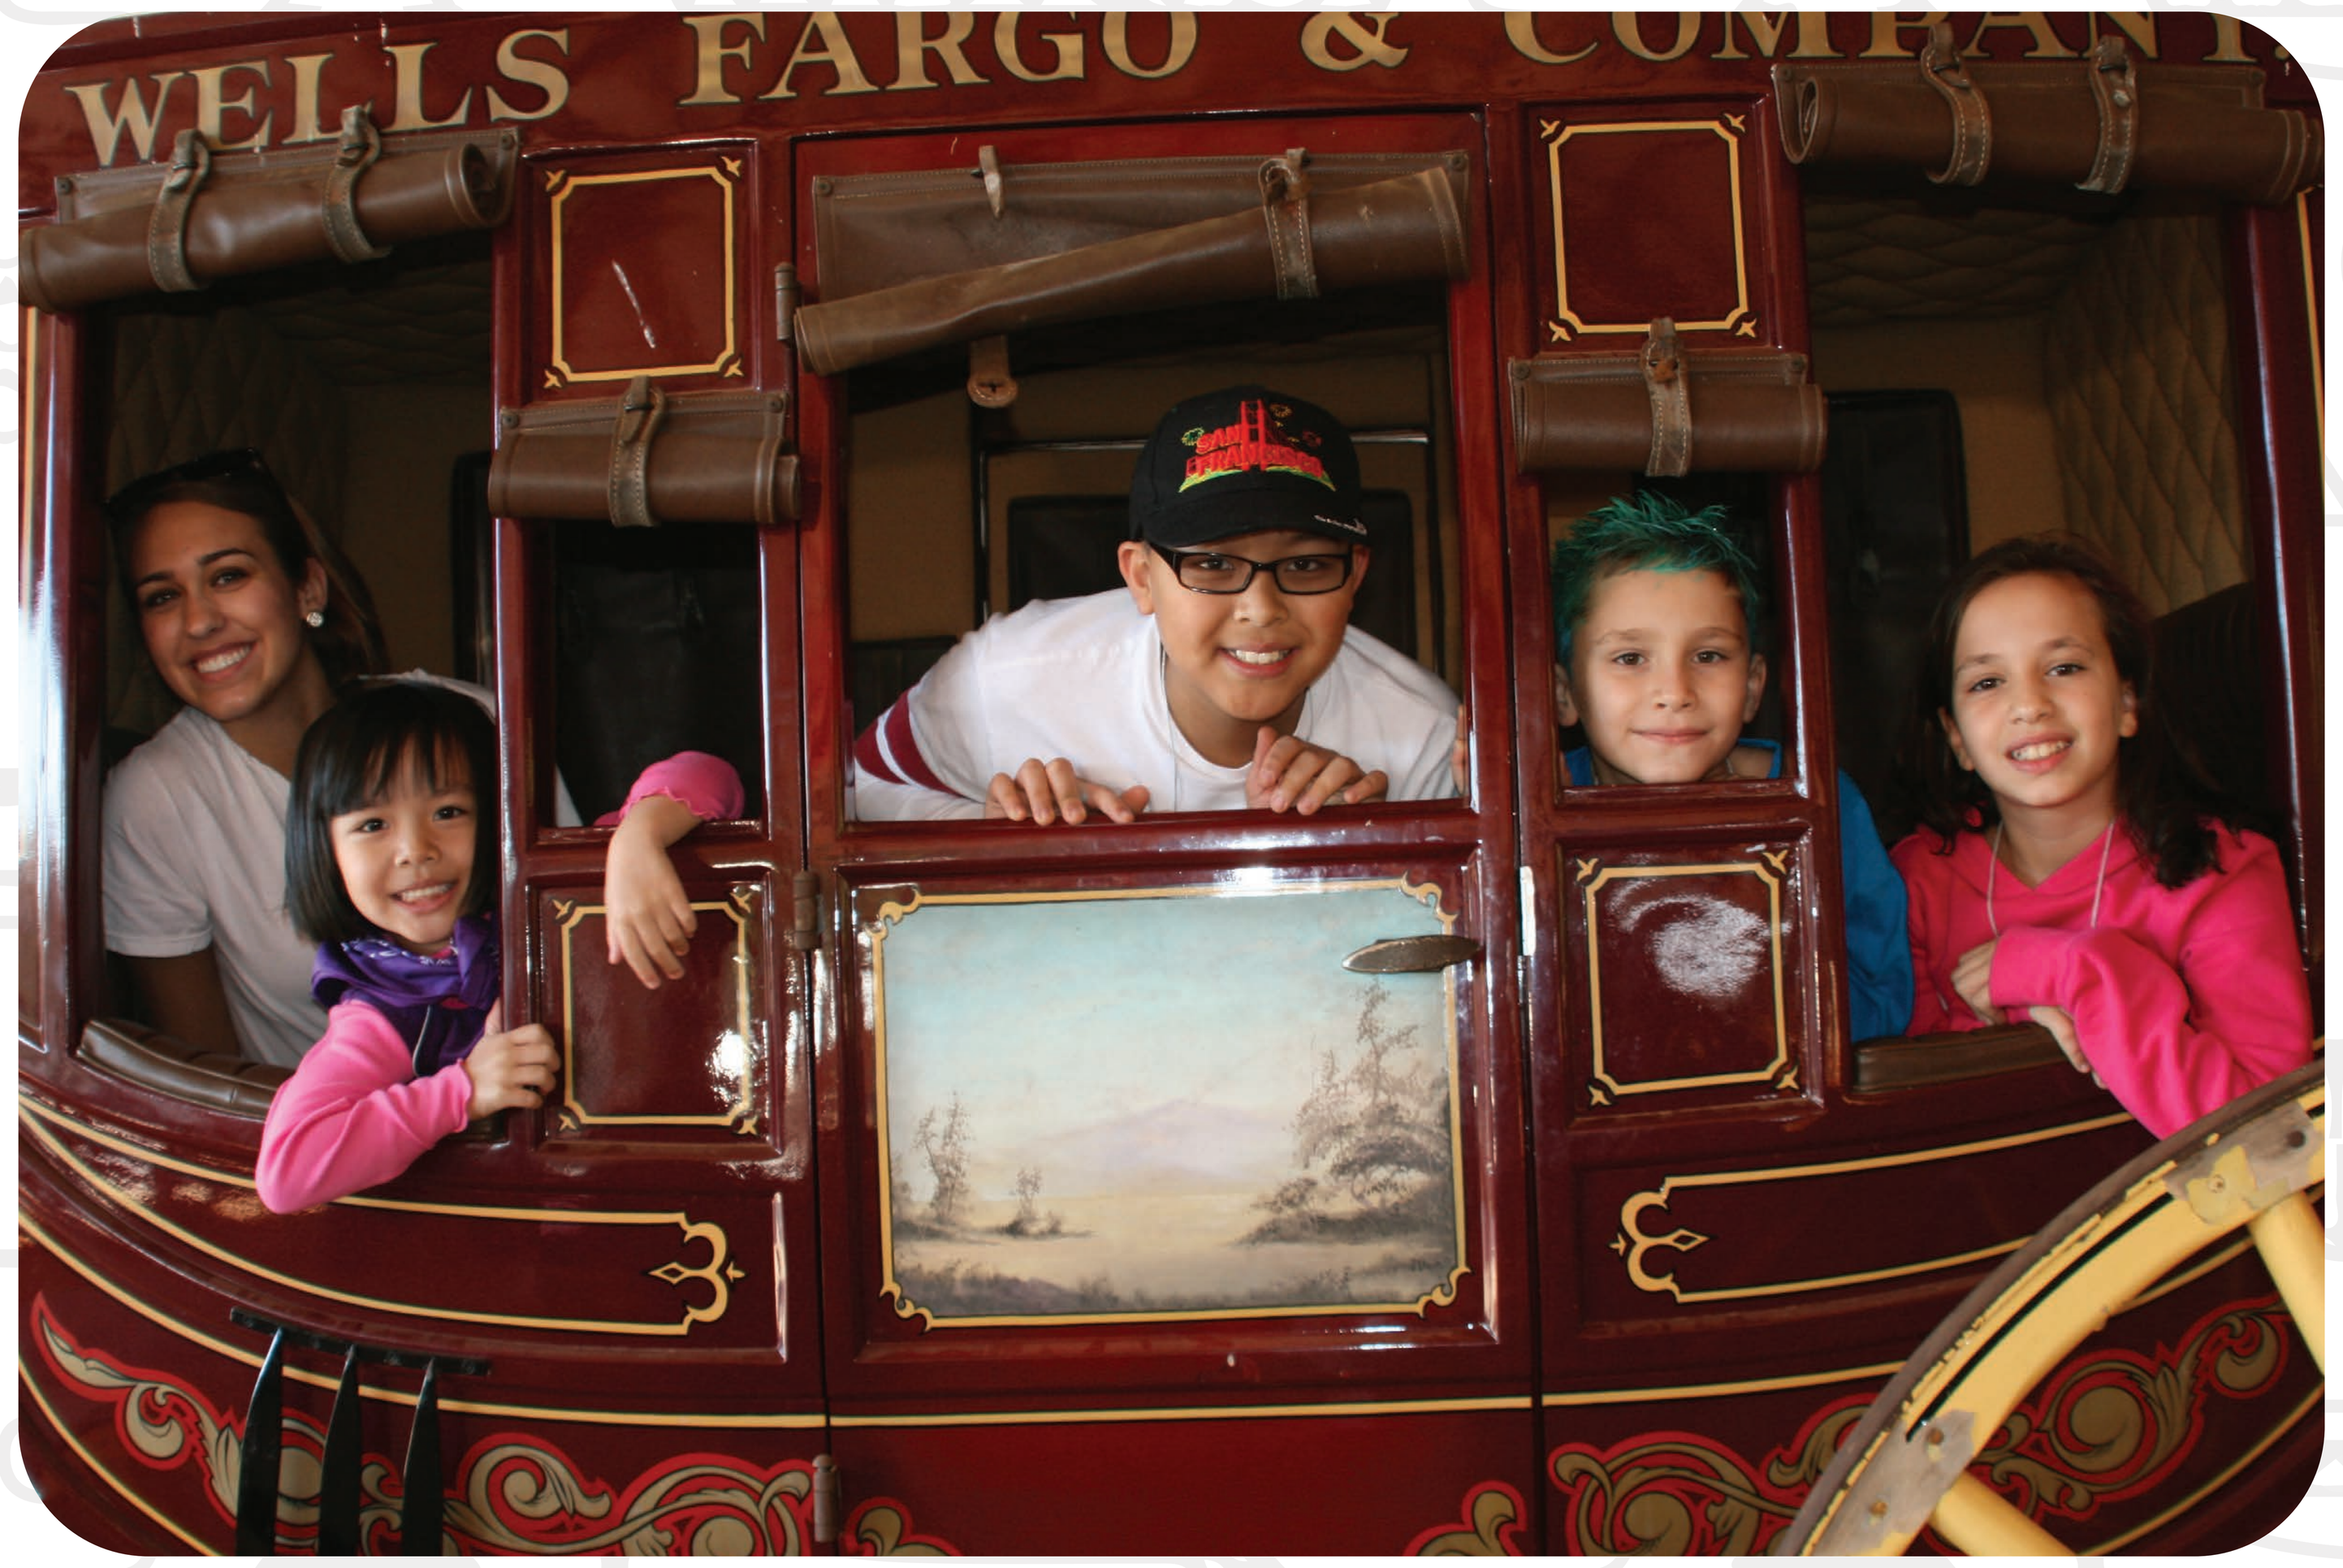
WELLS FARGO &

Materials and Methods

From 2010-2015 Comprehensive Health Education Services has provided a national program for children and their families affected by an active inhibitor. Many children with an inhibitor cannot attend annual hemophilia camp for a myriad of reasons; chief among them safety. IFC provides multiple opportunities for family fun and silliness in addition to educational programming for parents on the psychosocial effects of raising a child with an inhibitor.

To accomplish this we reached out to all national hemophilia chapters, hemophilia treatment centers and home health care companies, with a request to share information about IFC in newsletters, websites and social media. We distributed information via physical mailings and social media. We also requested that the national organizations include information regarding IFC in social media posts, newsletters and mailing lists.

Results

It quickly became evident that IFC would be their child's only annual camp experience as indicated by 55% of parents surveyed.

Since 2010, we've provided camp for residents in 33 of the 50 states.

Families report a 94% satisfaction rating with the program.

In 10 sessions, we served:

91 Families

193 Patients*

722 Individuals

Conclusions

IFC is an essential program for children and families affected by hemophilia and an inhibitor. Here at camp, our families become FAMILY for one another. The joy that is evident in each child's face as they find a friend who knows, TRULY knows what their struggles are, is difficult to put into words.

" Without this program my family would not have a chance to feel *normal*. My son would not have the chance to understand that he is not alone, there are other kids like him. It was life changing for us in sooo many ways"

- IFC FAMILY CAMPER

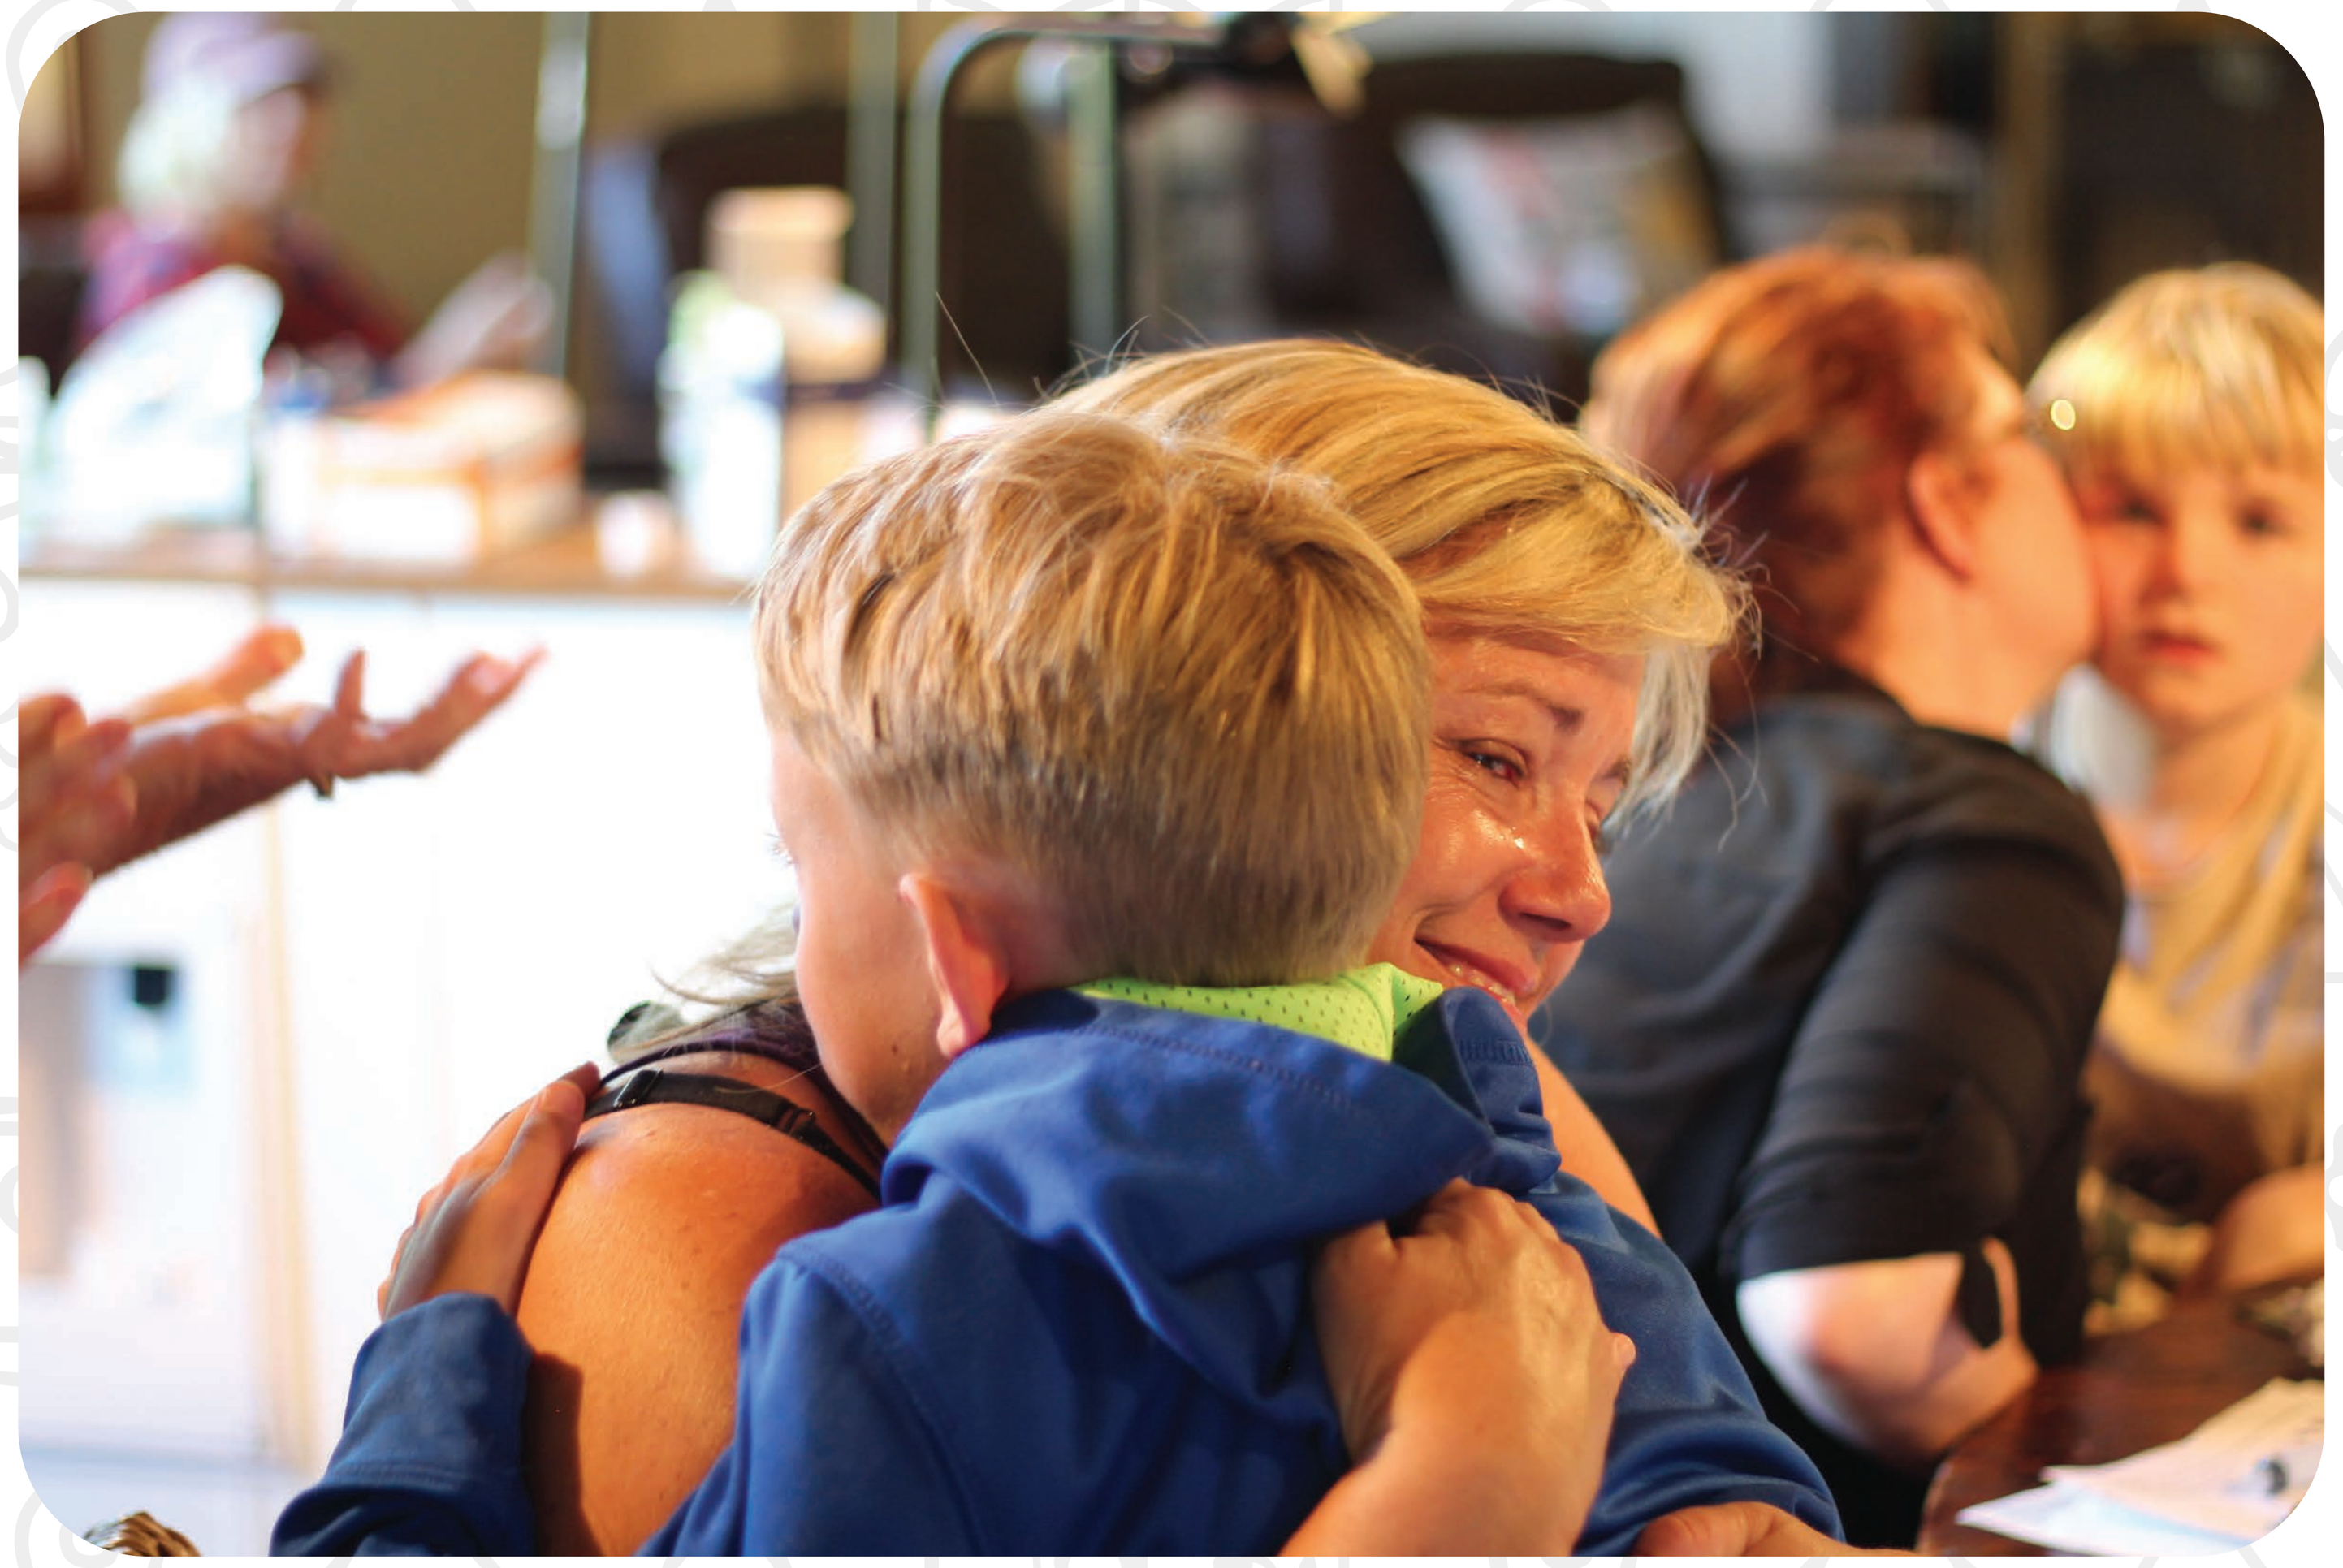

Conflicts of interest and all outside sources of funding

The author and owners of Comprehensive Health Education Services receive educational grants sponsored by Novo Nordisk, Baxalta, Grifols, Kedrion for their programming. IFC is supported by an educational grant from Novo Nordisk.

Eric Lowe and Janet Brewer have both been on consumer advisory boards for Novo Nordisk. Eric has also been featured in promotional materials developed by Novo Nordisk.

**Includes multiple attendances*

PosterSessionOnline.com

Poster Presented at: WFH 2016 World Congress

DOI: 10.3252/pso.eu.WFH2016.2016

Quality of Life
Janet Brewer
187--PP-M
WFH2016