MDS-157

Overall Survival, Clinical Benefit, and Durable Red Blood Cell Transfusion Independence With Imetelstat in the IMerge Phase 3 Trial of RBC Transfusion-Dependent Lower-Risk Myelodysplastic Syndromes

Valeria Santini,[1]* Rami S. Komrokji,[2]* Mikkael A. Sekeres,[3] Michael R. Savona,[4] Pierre Fenaux,[5] Yazan F. Madanat,[6] Esther Oliva,[7] Rena Buckstein,[8] Anna Jonášová,[9] Ulrich Germing,[10] Moshe Mittelman,[11] Sylvain Thépot,[12] Jennifer Riggs,[13] Souria Dougherty,[13] Tymara Berry,[13] Shyamala Navada,[13] Qi Xia,[13] Libo Sun,[13] Amer M. Zeidan,[14]* Uwe Platzbecker[15]*

[1]MDS Unit, Hematology, DMSC, University of Florence, AOUC, Florence, Italy; [2]Moffitt Cancer Center, Tampa, FL, USA; [3]Division of Hematology, Sylvester Comprehensive Cancer Center, University of Miami, Miami, FL, USA; [4]Vanderbilt-Ingram Cancer Center, Vanderbilt University Medical Center, Nashville, TN, USA; [5]Service d'Hématologie Séniors, Hôpital Saint-Louis, Université de Paris 7, Paris, France; [6]Harold C. Simmons Comprehensive Cancer Center, UT Southwestern Medical Center, Dallas, TX, USA; [7]Grand Metropolitan Hospital, Reggio Calabria, Italy; [8]Odette Cancer Centre, Sunnybrook Health Sciences Centre, Toronto, ON, Canada; [9]Department of Medicine – Department of Hematology of the 1st Faculty of Medicine and General University Hospital, Prague, Czech Republic; [10]Universitätsklinikum Düsseldorf Hämatologie, Onkologie, Düsseldorf, Germany; [11]Tel Aviv Sourasky Medical Center, Tel Aviv, Israel; [12]Centre Hospitalier Universitaire d'Angers, Angers, France; [13]Geron Corporation, Foster City, CA, USA; [14]Yale School of Medicine and Yale Cancer Center, Yale University, New Haven, CT, USA; [15]Department of Hematology, Cellular Therapy and Hemostaseology, Leipzig University Hospital, Leipzig, Germany

*Authors contributed equally

Background

- Current treatments for lower-risk myelodysplastic syndromes (LR-MDS) have limitations, and patients often become red blood cell (RBC) transfusion-dependent (TD)[1,2]
- RBC transfusions negatively affect progression-free survival (PFS) and overall survival (OS) in patients with MDS, highlighting the need for therapies that sustain continuous RBC-transfusion independence (TI) and have the potential to improve OS[1,2]
- Imetelstat is a recently United States Food and Drug Administration approved, first-in-class, direct, and competitive inhibitor of telomerase enzymatic activity that selectively targets malignant clones and allows for recovery of erythropoiesis[3-5]
- IMerge (NCT02598661) is a multinational, randomized, double-blind, phase 2/3 trial of imetelstat versus placebo in patients with heavily RBC-TD, erythropoiesis-stimulating agent (ESA)–relapsed or –refractory (R/R) or ineligible, and non-del(5q) LR-MDS who were naive to treatment with lenalidomide or hypomethylating agents[6]
 - Phase 3 primary analysis demonstrated significantly higher rates of ≥8-week, ≥24-week, and ≥1-year RBC-TI with imetelstat (n=118; 39.8%, 28.0%, and 17.8%, respectively) versus placebo (n=60; 15.0%, 3.3%, and 1.7%, respectively) in the intention-to-treat population
 - Most common adverse events were neutropenia and thrombocytopenia, which were generally resolvable to grade 2 and below and manageable with dose modifications
- This analysis assessed OS and clinical benefit of durable RBC-TI as the secondary end points in IMerge

Methods

Study design

- IMerge was a double-blind, randomized (2:1), placebo-controlled, phase 3 clinical trial conducted at 118 global sites that enrolled patients between 2019 and 2022 (**Figure 1**)
- The primary end point was ≥8-week RBC-TI rate; secondary end points included ≥24-week RBC-TI, duration of RBC-TI, hematologic improvement-erythroid (HI-E), OS, and safety
- Primary analysis cutoff was October 2022, with October 2023 cutoff for ≥1-year RBC-TI analyses, and January 2024 for OS

Patients

- Enrollment included patients with RBC-TD (defined as requiring ≥4 RBC U/8 weeks), ESA-R/R or ineligible non-del(5q) LR-MDS naïve to lenalidomide and hypomethylating agents with low-risk or intermediate-1–risk disease per International Prognostic Scoring System (IPSS)

Statistics

- RBC-TI duration and OS were calculated by the Kaplan-Meier method and compared via the stratified log-rank test

Figure 1. IMerge phase 3 study design

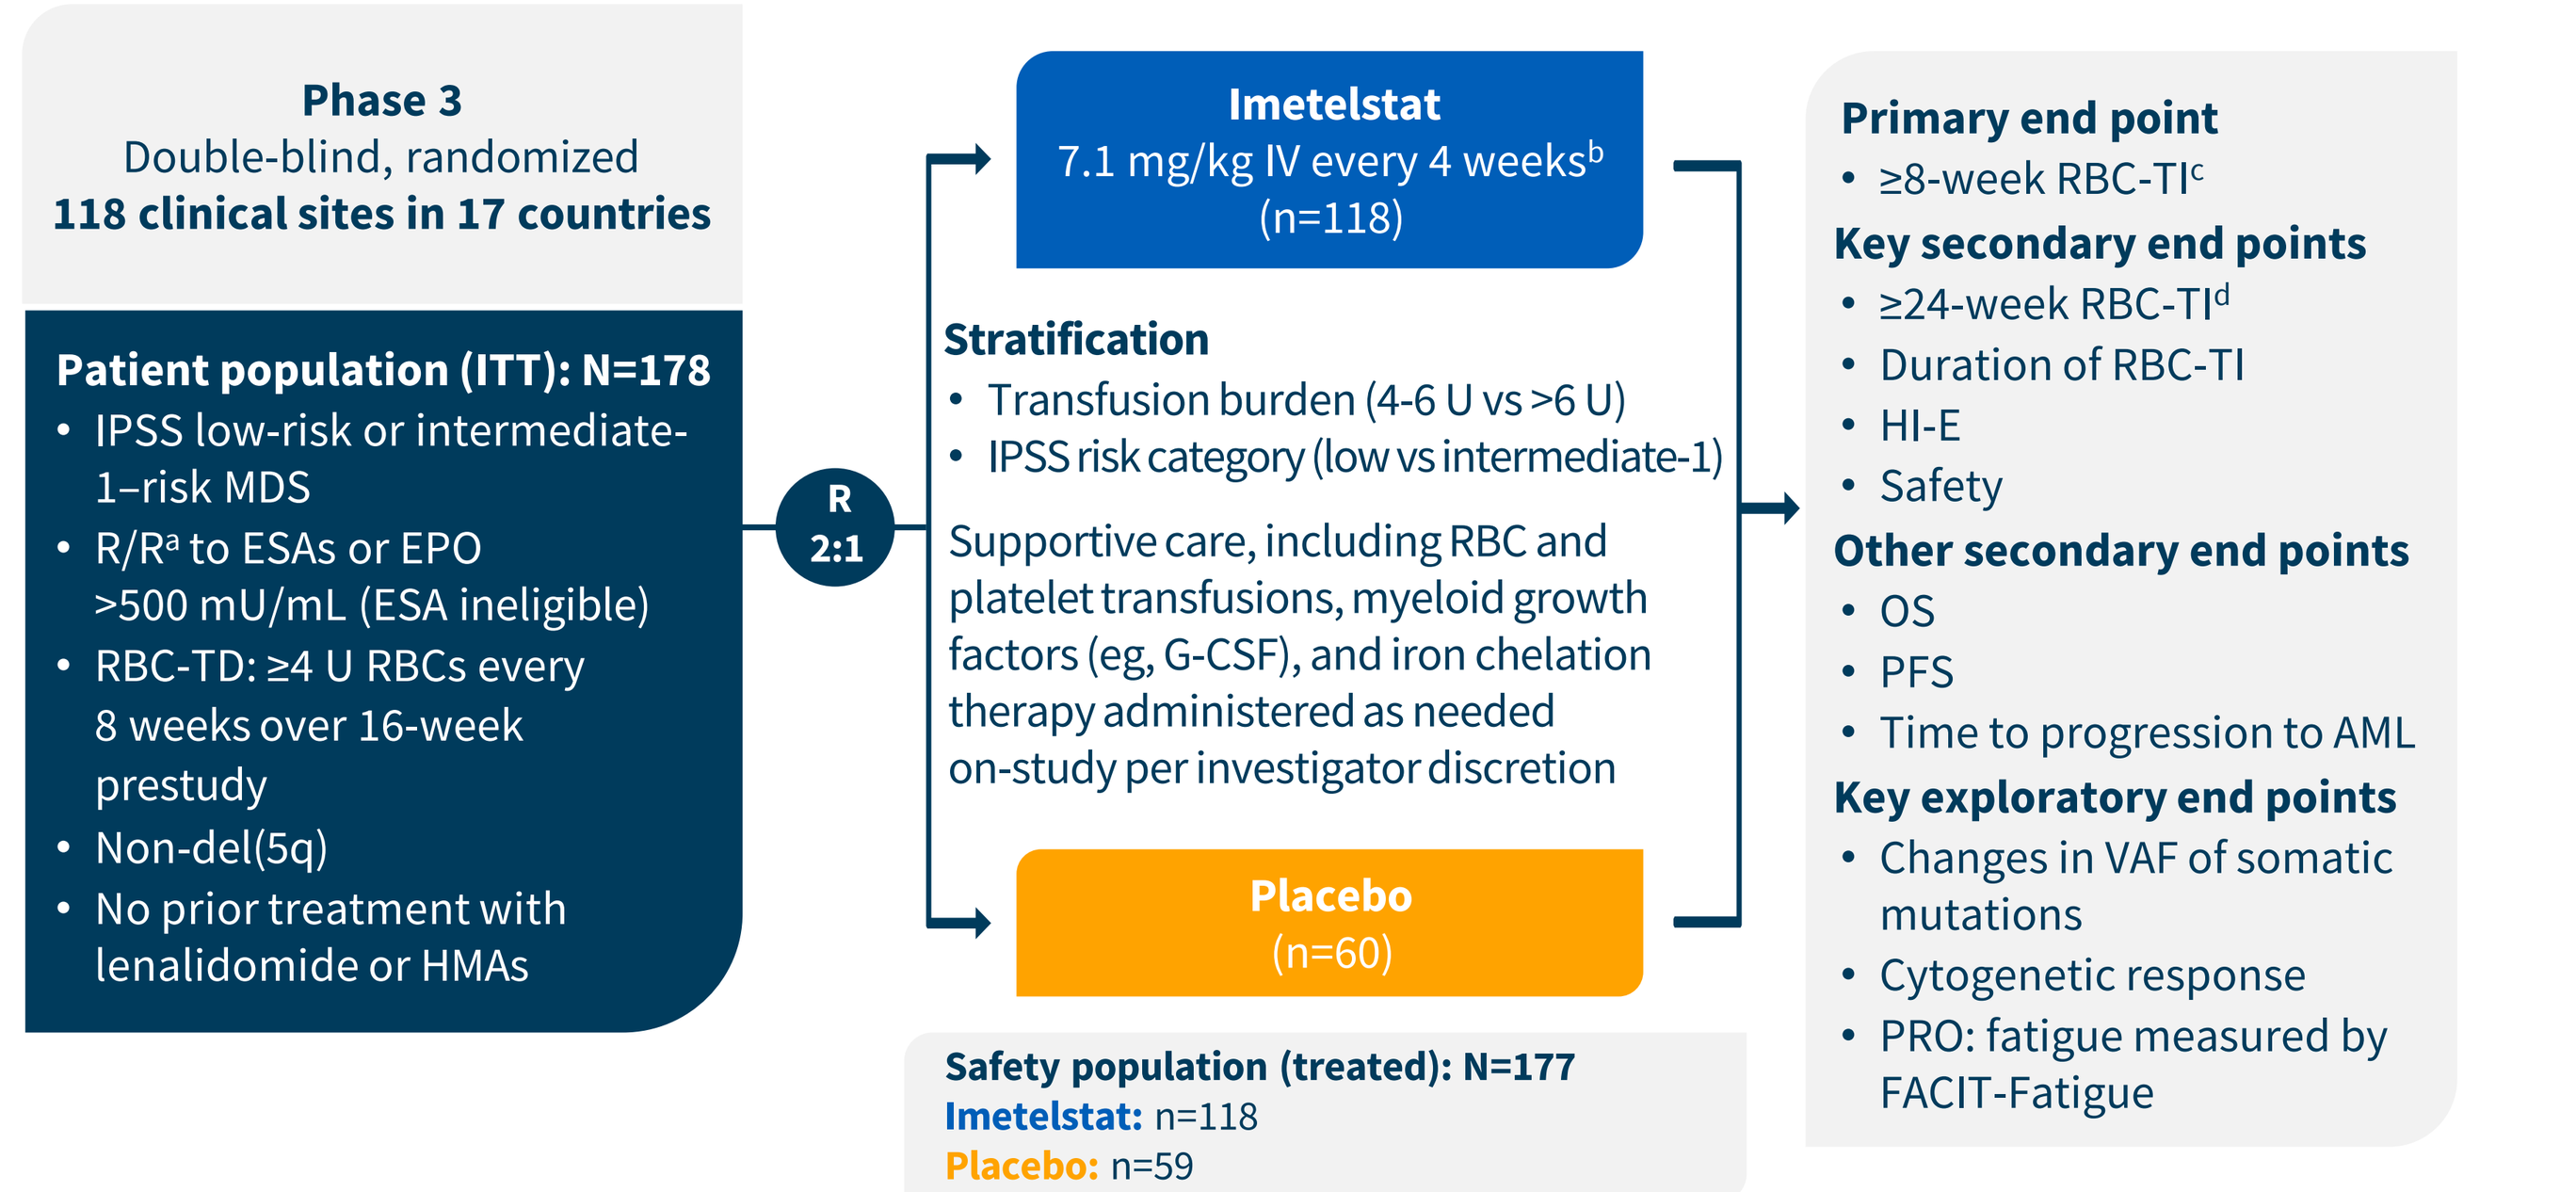
Phase 3
Double-blind, randomized
118 clinical sites in 17 countries
Patient population (ITT): N=178
IPSS low-risk or intermediate-1–risk MDS
R/Rᵃ to ESAs or EPO >500 mU/mL (ESA ineligible)
RBC-TD: ≥4 U RBCs every 8 weeks over 16-week prestudy
Non-del(5q)
No prior treatment with lenalidomide or HMAs
R 2:1
Imetelstat
7.1 mg/kg IV every 4 weeksᵇ
(n=118)
Stratification
Transfusion burden (4-6 U vs >6 U)
IPSS risk category (low vs intermediate-1)
Supportive care, including RBC and platelet transfusions, myeloid growth factors (eg, G-CSF), and iron chelation therapy administered as needed on-study per investigator discretion
Placebo
(n=60)
Safety population (treated): N=177
Imetelstat: n=118
Placebo: n=59
Primary end point
≥8-week RBC-TIᶜ
Key secondary end points
≥24-week RBC-TIᵈ
Duration of RBC-TI
HI-E
Safety
Other secondary end points
OS
PFS
Time to progression to AML
Key exploratory end points
Changes in VAF of somatic mutations
Cytogenetic response
PRO: fatigue measured by FACIT-Fatigue

[a]Received ≥8 weeks of ESA treatment (epoetin alfa ≥40,000 U, epoetin beta ≥30,000 U, or darbepoetin alfa 150 µg or equivalent per week) without Hb rise ≥1.5 g/dL or decreased RBC transfusion requirement ≥4 U every 8 weeks or transfusion dependence or reduction in Hb by ≥1.5 g/dL after hematologic improvement from ≥8 weeks of ESA treatment. [b]Equivalent to 7.5 mg/kg imetelstat sodium. [c]Proportion of patients without any RBC transfusion for ≥8 consecutive weeks since entry to the trial (≥8-week RBC-TI). [d]Proportion of patients without any RBC transfusion for ≥24 consecutive weeks since entry to the trial (≥24-week RBC-TI).
AML, acute myeloid leukemia; EPO, erythropoietin; ESA, erythropoiesis-stimulating agent; FACIT, Functional Assessment of Chronic Illness Therapy; G-CSF, granulocyte colony-stimulating factor; Hb, hemoglobin; HI-E, hematologic improvement-erythroid; HMA, hypomethylating agent; IPSS, International Prognostic Scoring System; ITT, intention-to-treat; IV, intravenous; MDS, myelodysplastic syndromes; OS, overall survival; PFS, progression-free survival; PRO, patient-reported outcome; RBC, red blood cell; R/R, relapsed or refractory; TD, transfusion dependent; TI, transfusion independence; VAF, variant allele frequency.

Results

- Baseline demographic and disease characteristics were similar across treatment arms (**Table 1**)

Table 1. Baseline Characteristics

Baseline demographic and disease characteristics	Imetelstat (n=118)	Placebo (n=60)
Median (range) age, y	72 (44-87)	73 (39-85)
Male, n (%)	71 (60)	40 (67)
Median (range) time since diagnosis, y	3.5 (0.1-26.7)	2.8 (0.2-25.7)
WHO classification, n (%)		
RS+	73 (62)	37 (62)
RS–	44 (37)	23 (38)
IPSS risk category, n (%)		
Low	80 (68)	39 (65)
Intermediate-1	38 (32)	21 (35)
Median (range) pretreatment Hb,[a] g/dL	7.9 (5.3-10.1)	7.8 (6.1-9.2)
Median (range) prior RBC transfusion burden, RBC U/8 weeks	6 (4-33)	6 (4-13)
Prior RBC transfusion burden, n (%)		
≥4 to ≤6 U/8 weeks	62 (53)	33 (55)
>6 U/8 weeks	56 (48)	27 (45)
Median (range) sEPO, mU/mL	174.9 (6.0-4460.0)	277.0 (16.9-5514.0)
sEPO level, n (%)[b]		
≤500 mU/mL	87 (74)	36 (60)
>500 mU/mL	26 (22)	22 (37)
Prior ESA, n (%)	108 (92)	52 (87)
Prior luspatercept, n (%)[c]	7 (6)	4 (7)

Data cutoff date: October 2022. [a]Average of all Hb values in the 8 weeks before the first dose date, excluding values within 14 days after a transfusion, which was considered to be influenced by transfusion. [b]Data missing for 5 patients in the imetelstat group and 2 in the placebo group. [c]Insufficient number of patients previously treated with luspatercept to draw conclusions about the effect of imetelstat treatment in such patients.
Hb, hemoglobin; ESA, erythropoiesis-stimulating agent; IPSS, International Prognostic Scoring System; RBC, red blood cell; RS, ring sideroblast; sEPO, serum erythropoietin; WHO, World Health Organization.

- Of imetelstat responders (n=47), 83% had a single, continuous RBC-TI period (**Figure 2**)
- Median duration of RBC-TI in imetelstat responders was longer (51.6 weeks) than with placebo (n=9; 13.3 weeks)

Figure 2. Continuous RBC-TI Periods in ≥8-Week RBC-TI Responders

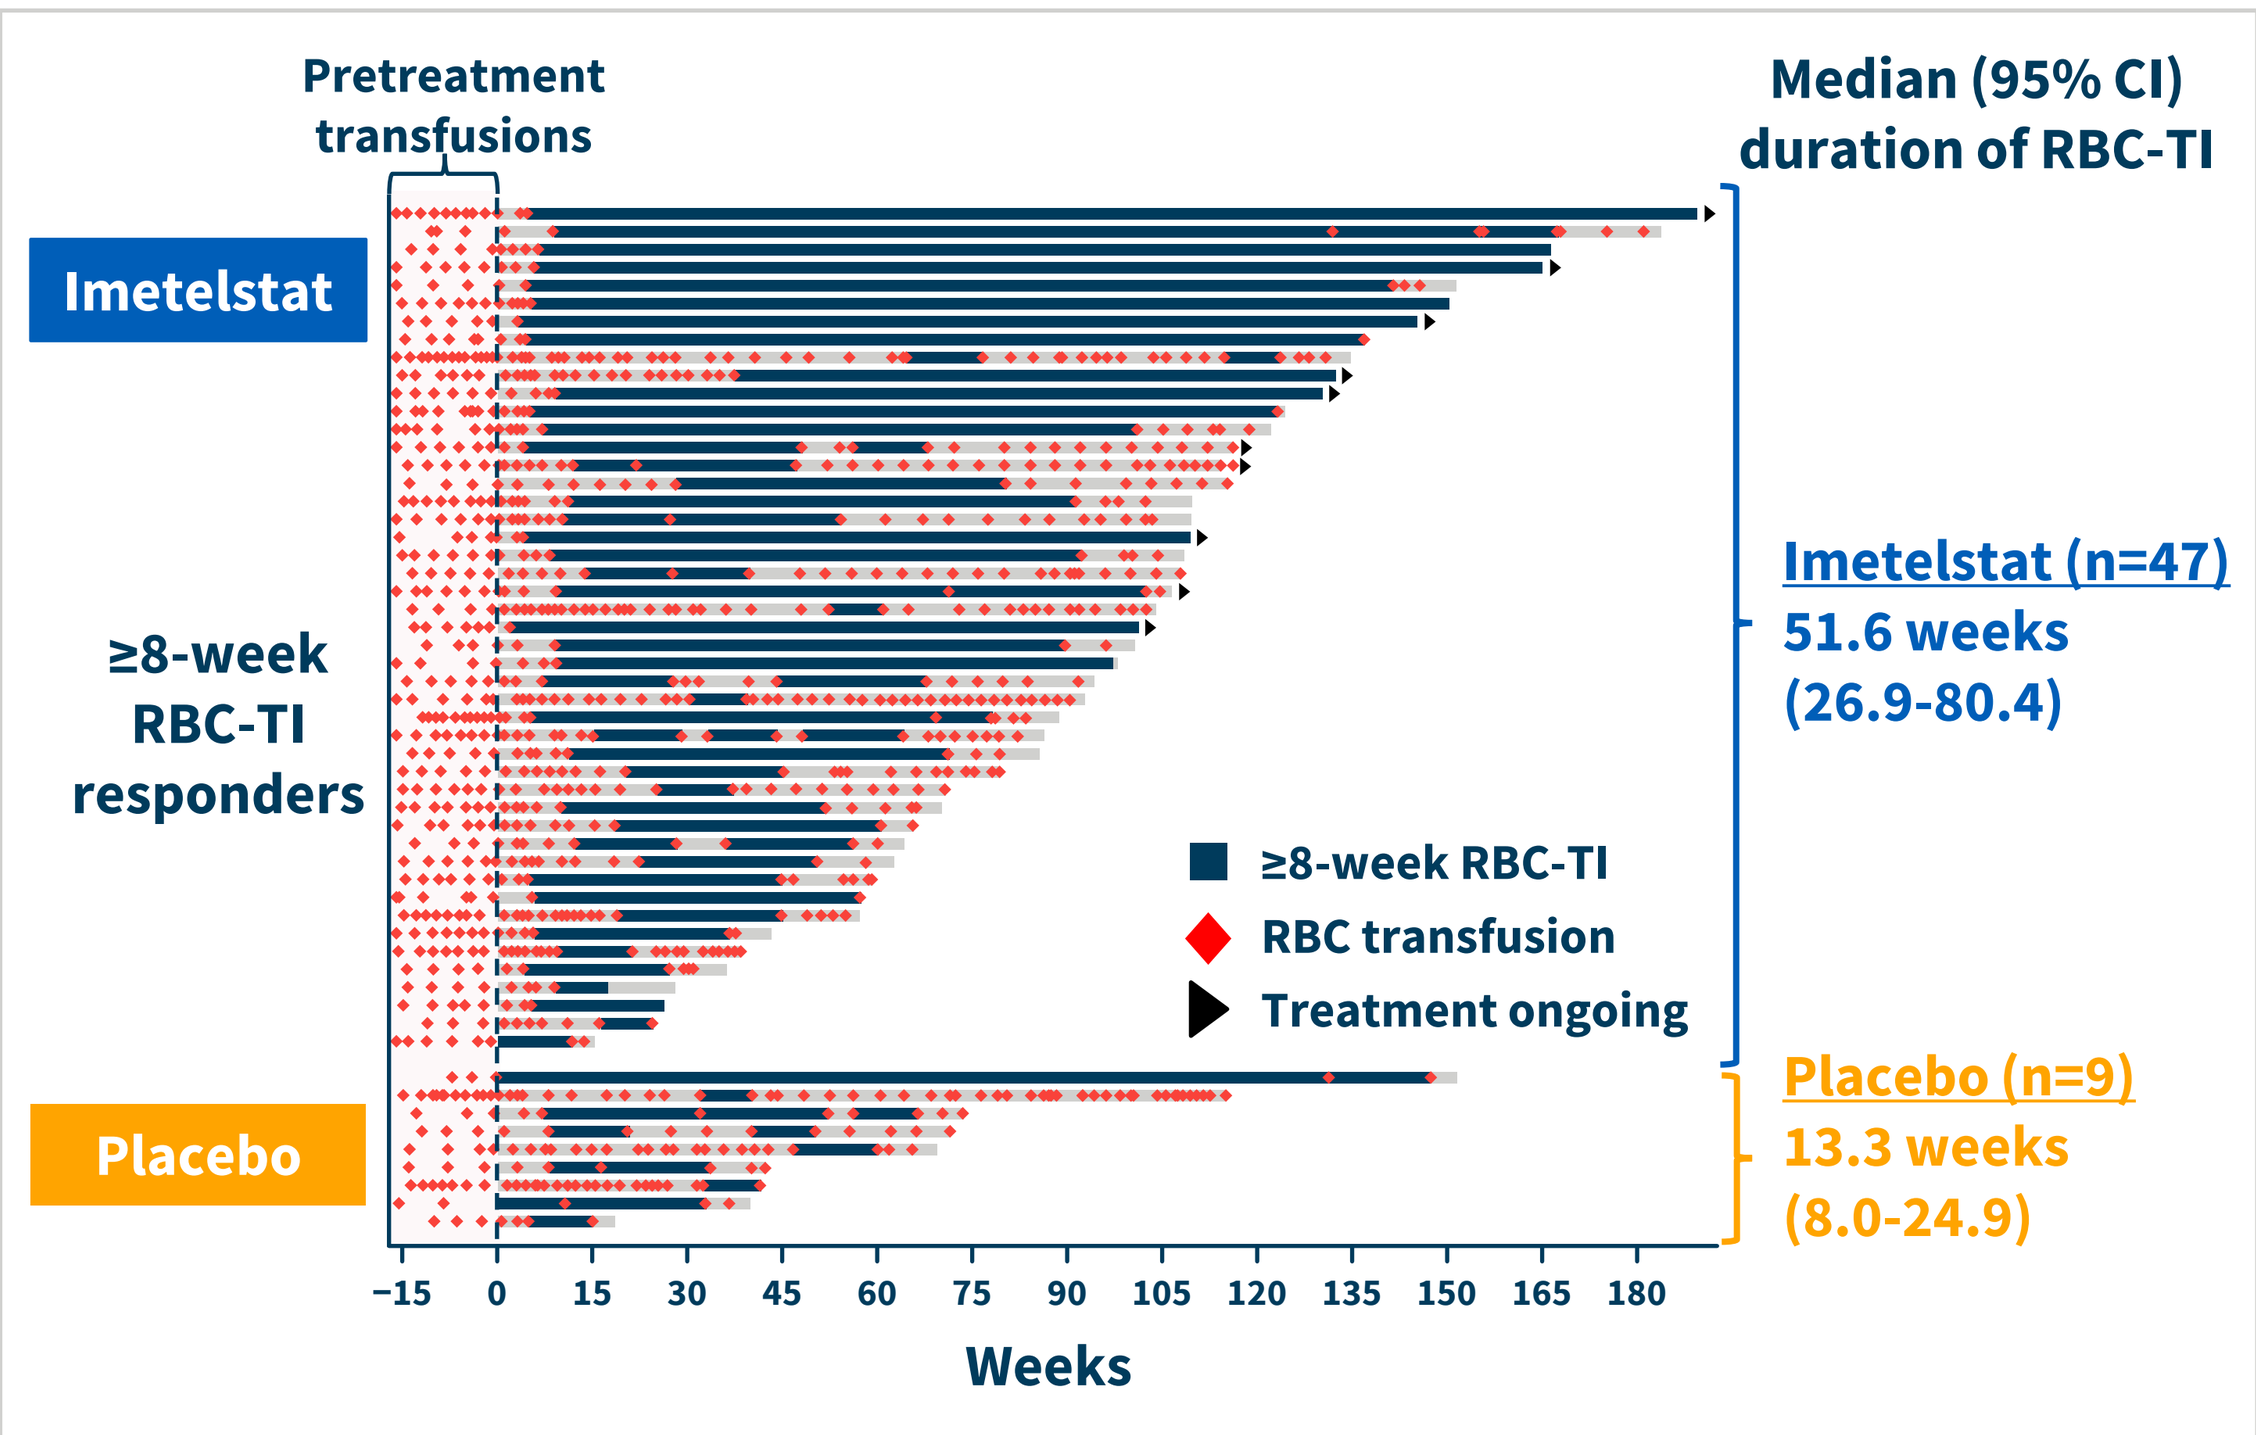
Pretreatment transfusions
Median (95% CI) duration of RBC-TI
Imetelstat
≥8-week RBC-TI responders
Placebo
≥8-week RBC-TI
RBC transfusion
Treatment ongoing
Imetelstat (n=47)
51.6 weeks
(26.9-80.4)
Placebo (n=9)
13.3 weeks
(8.0-24.9)
−15 0 15 30 45 60 75 90 105 120 135 150 165 180
Weeks

Data cutoff date: October 2023.
RBC, red blood cell; TI, transfusion independence.

- Among imetelstat responders, RBC-TI was durable and sustained over time (**Figure 3**)
 - Median RBC-TI duration for the imetelstat ≥8-week, ≥24-week, and ≥1-year RBC-TI responders was 52, 80, and 132 weeks, respectively
 - 70% of ≥8-week RBC-TI responders achieved ≥24-week RBC-TI response
 - 64% of ≥24-week RBC-TI responders achieved ≥1-year RBC-TI response

Figure 3. Achievement and Durability of RBC-TI

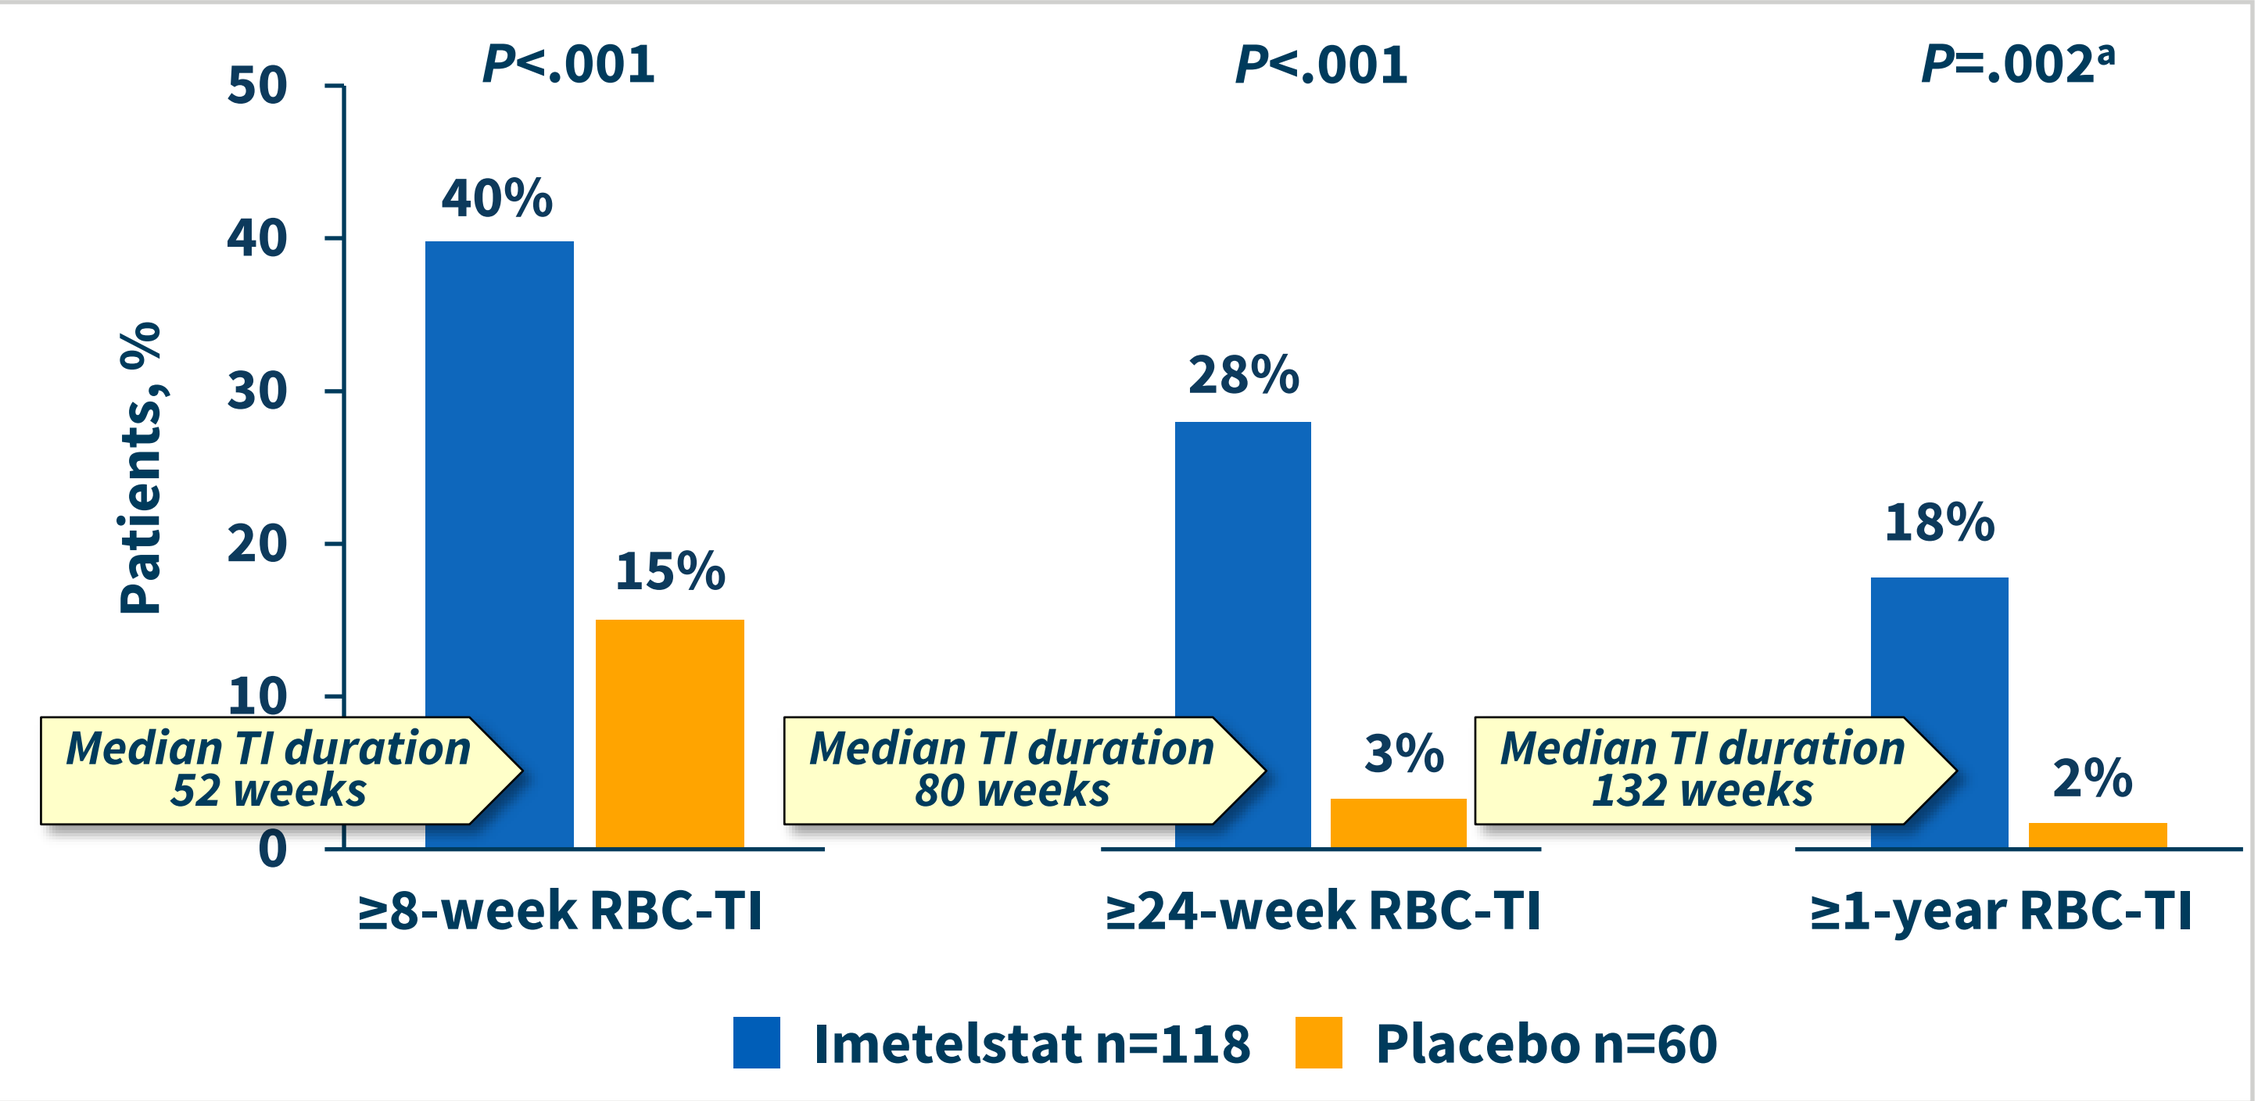
Patients, %
50 40 30 20 10 0
P<.001
P<.001
P=.002ᵃ
40%
15%
28%
3%
18%
2%
Median TI duration 52 weeks
Median TI duration 80 weeks
Median TI duration 132 weeks
≥8-week RBC-TI
≥24-week RBC-TI
≥1-year RBC-TI
Imetelstat n=118
Placebo n=60

[a]Reported as descriptive *P* value.
Data cutoff date: October 2023.
RBC, red blood cell; TI, transfusion independence.

- In an exploratory analysis of the total imetelstat-treated population, significantly more patients achieved RBC-TI with concurrent Hb increase ≥1.5 g/dL (**Figure 4**)
- Imetelstat responders had higher central Hb peaks versus placebo responders (**Figure 5**)
- There was no apparent detriment with imetelstat treatment on OS (**Figure 6**)
 - As of January 2024, 55 (47%) and 26 (43%) patients were in follow-up in the imetelstat and placebo groups, respectively
 - Median duration of follow-up was 32 months for imetelstat and 28 months for placebo
 - Two-year OS rates were 78% in the imetelstat group (81% in ≥8-week RBC-TI responders and 75% in nonresponders) versus 74% in the placebo group

Figure 4. Achievement of RBC-TI With Concurrent Hb Increase ≥1.5 g/dL

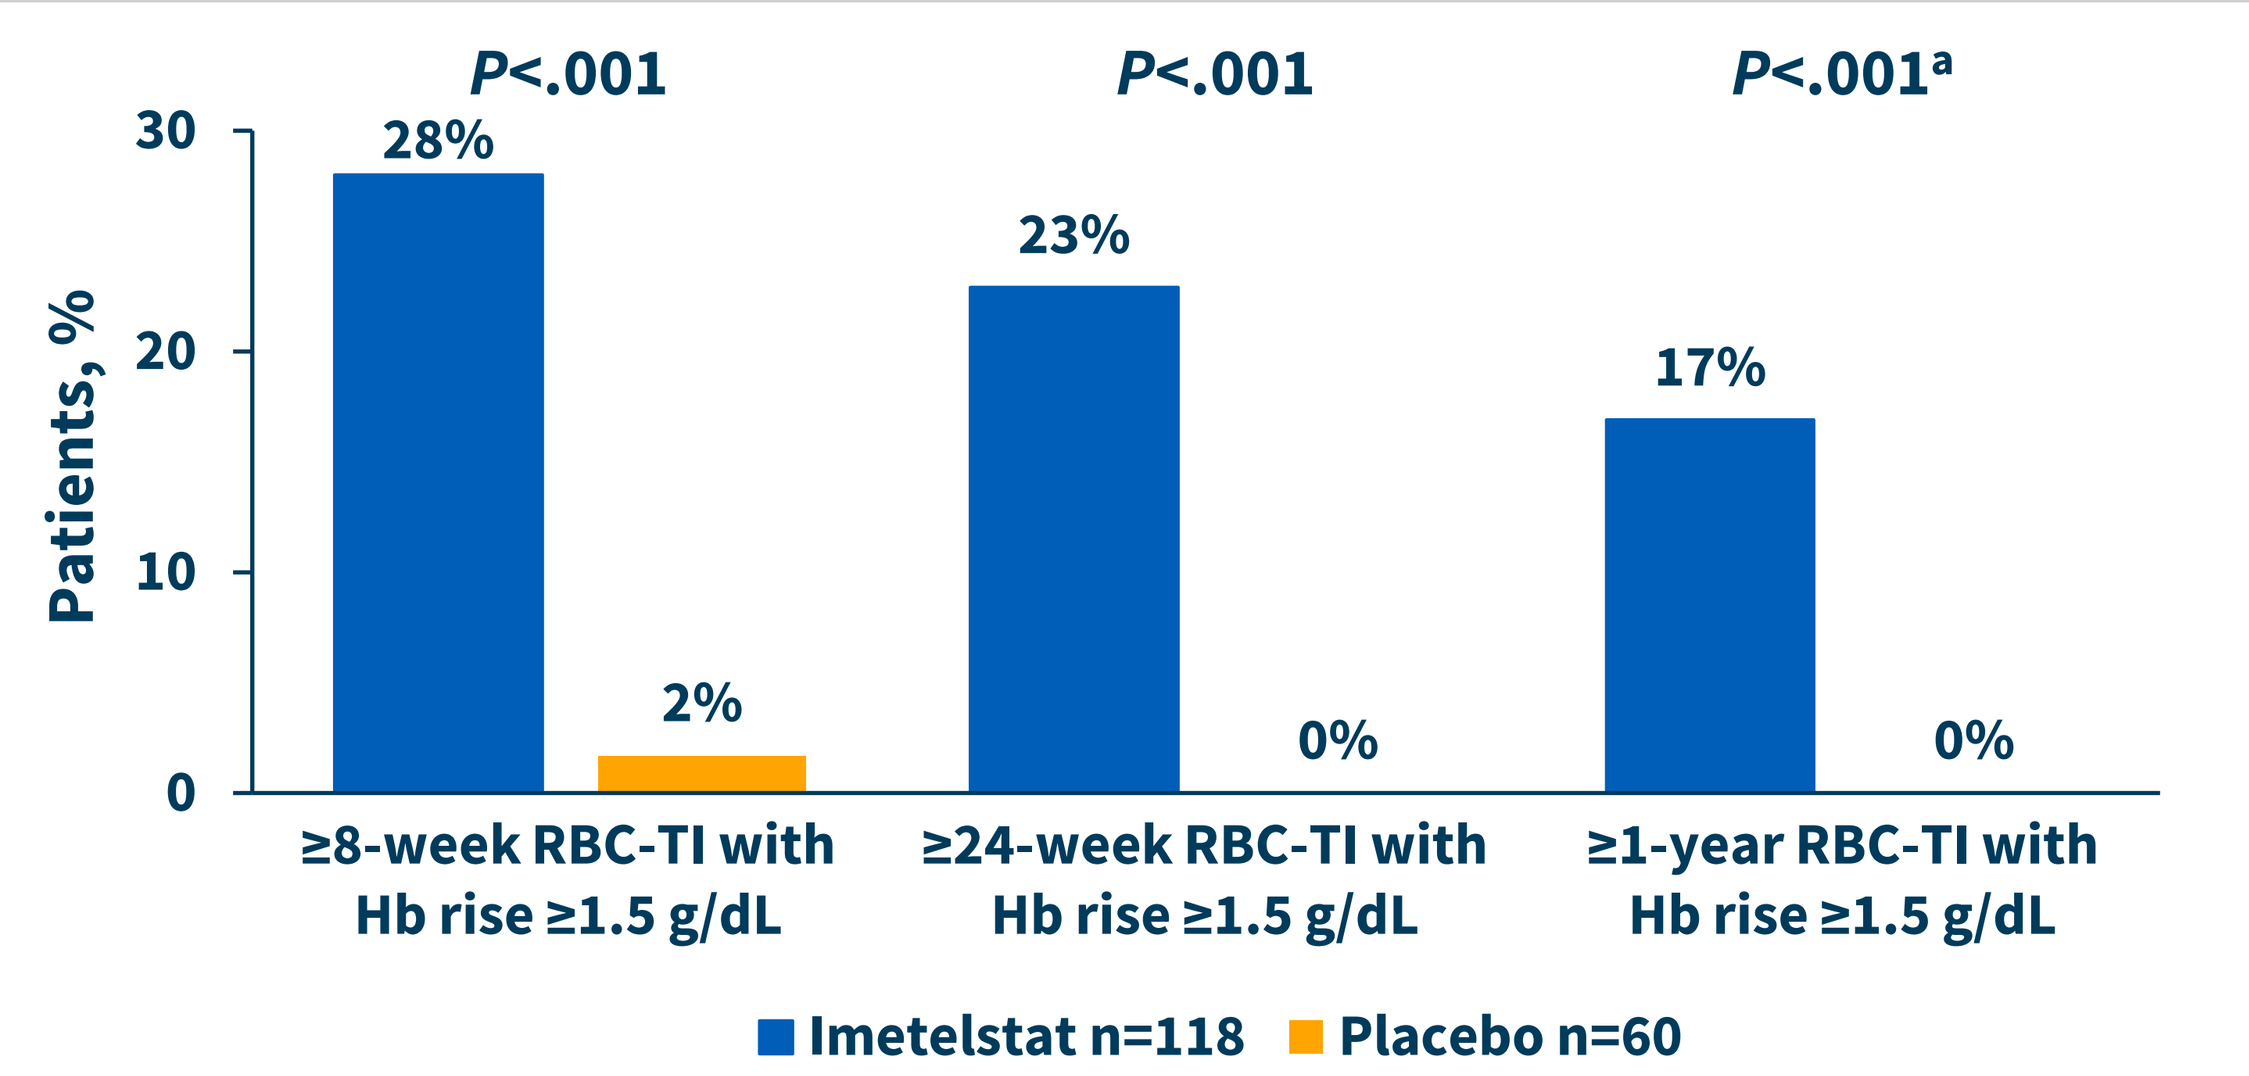
Patients, %
30 20 10 0
P<.001
P<.001
P<.001ᵃ
28%
2%
23%
0%
17%
0%
≥8-week RBC-TI with Hb rise ≥1.5 g/dL
≥24-week RBC-TI with Hb rise ≥1.5 g/dL
≥1-year RBC-TI with Hb rise ≥1.5 g/dL
Imetelstat n=118
Placebo n=60

Exploratory analysis.
[a]Reported as descriptive *P* value.
Data cutoff date: October 2023.
Hb, hemoglobin; RBC, red blood cell; TI, transfusion independence.

Figure 5. Median Hb Peak in RBC-TI Responders

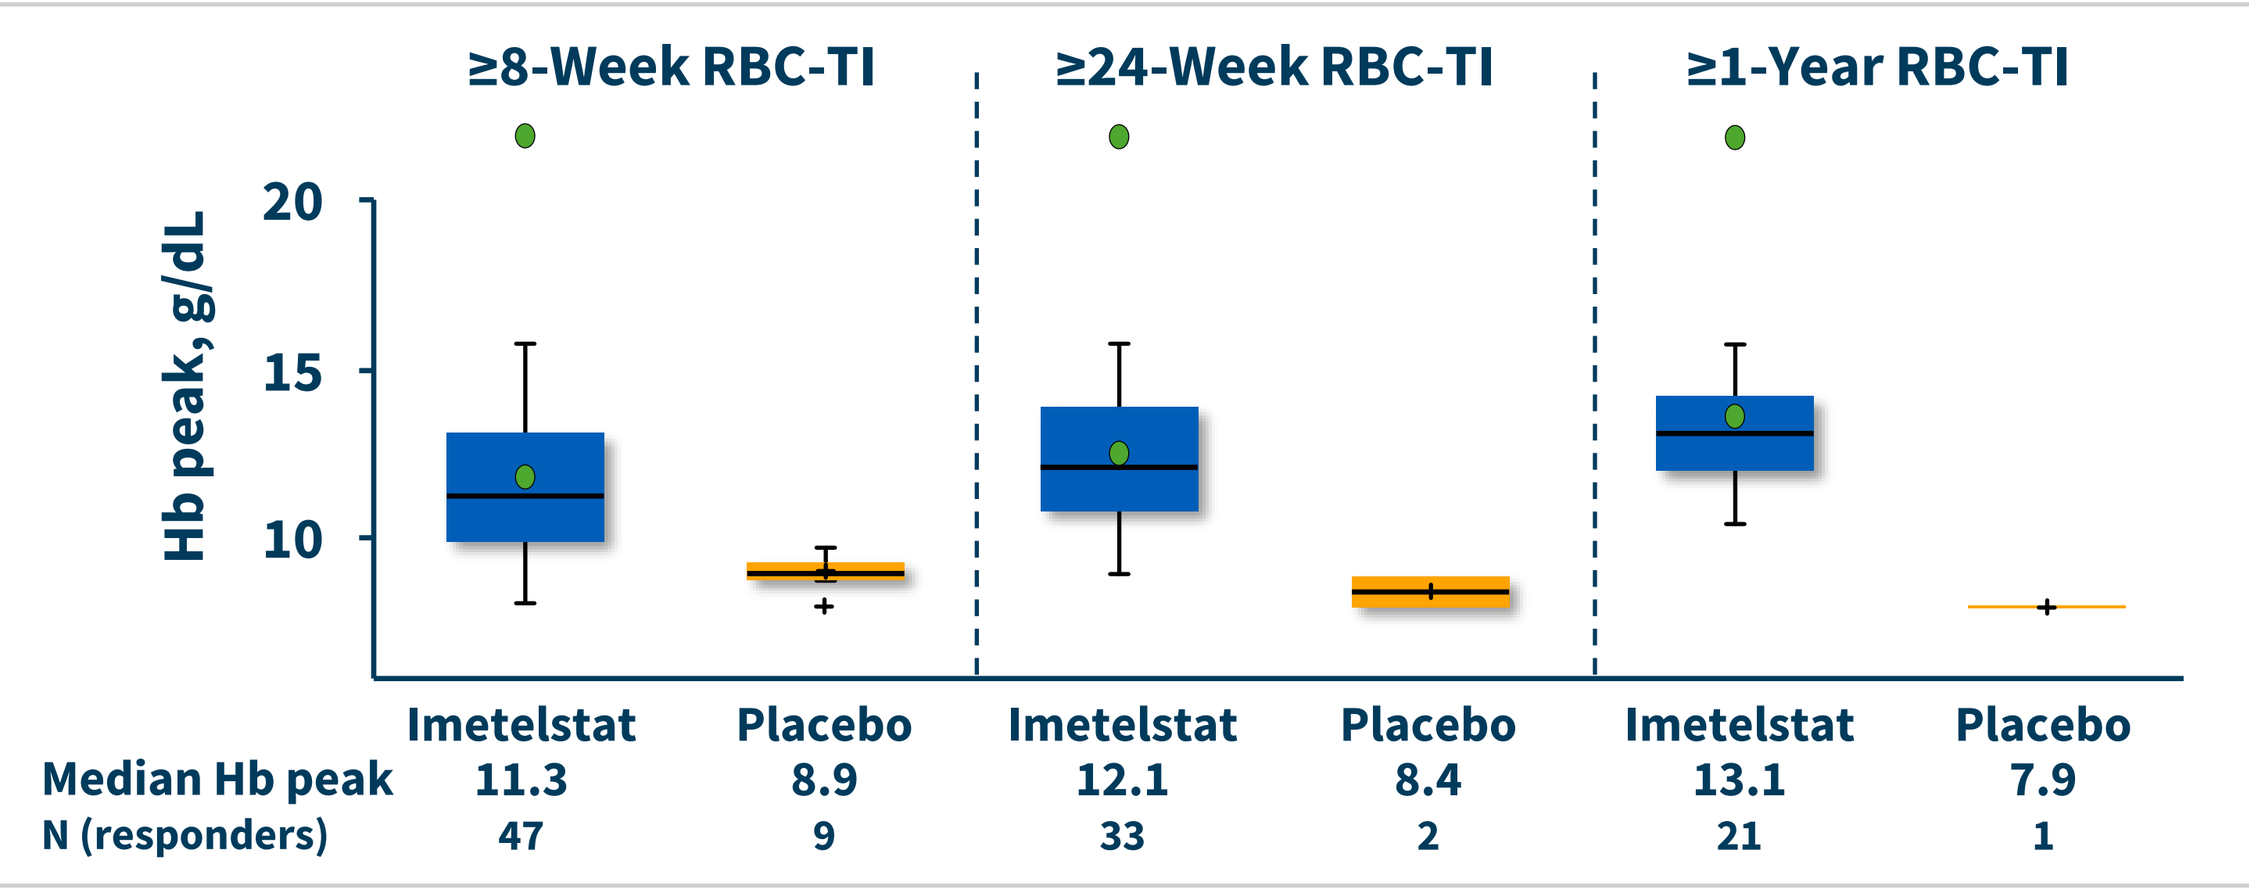
≥8-Week RBC-TI
≥24-Week RBC-TI
≥1-Year RBC-TI
Hb peak, g/dL
20 15 10
Imetelstat Placebo Imetelstat Placebo Imetelstat Placebo
Median Hb peak 11.3 8.9 12.1 8.4 13.1 7.9
N (responders) 47 9 33 2 21 1

Exploratory analysis. Hb peak is the maximum Hb value in the longest transfusion free interval excluding the first 2 weeks.
Data cutoff date: October 2023.
Hb, hemoglobin; RBC, red blood cell; TI, transfusion independence.

Figure 6. OS in IMerge for (A) Imetelstat Versus Placebo and (B) Imetelstat Responders Versus Nonresponders

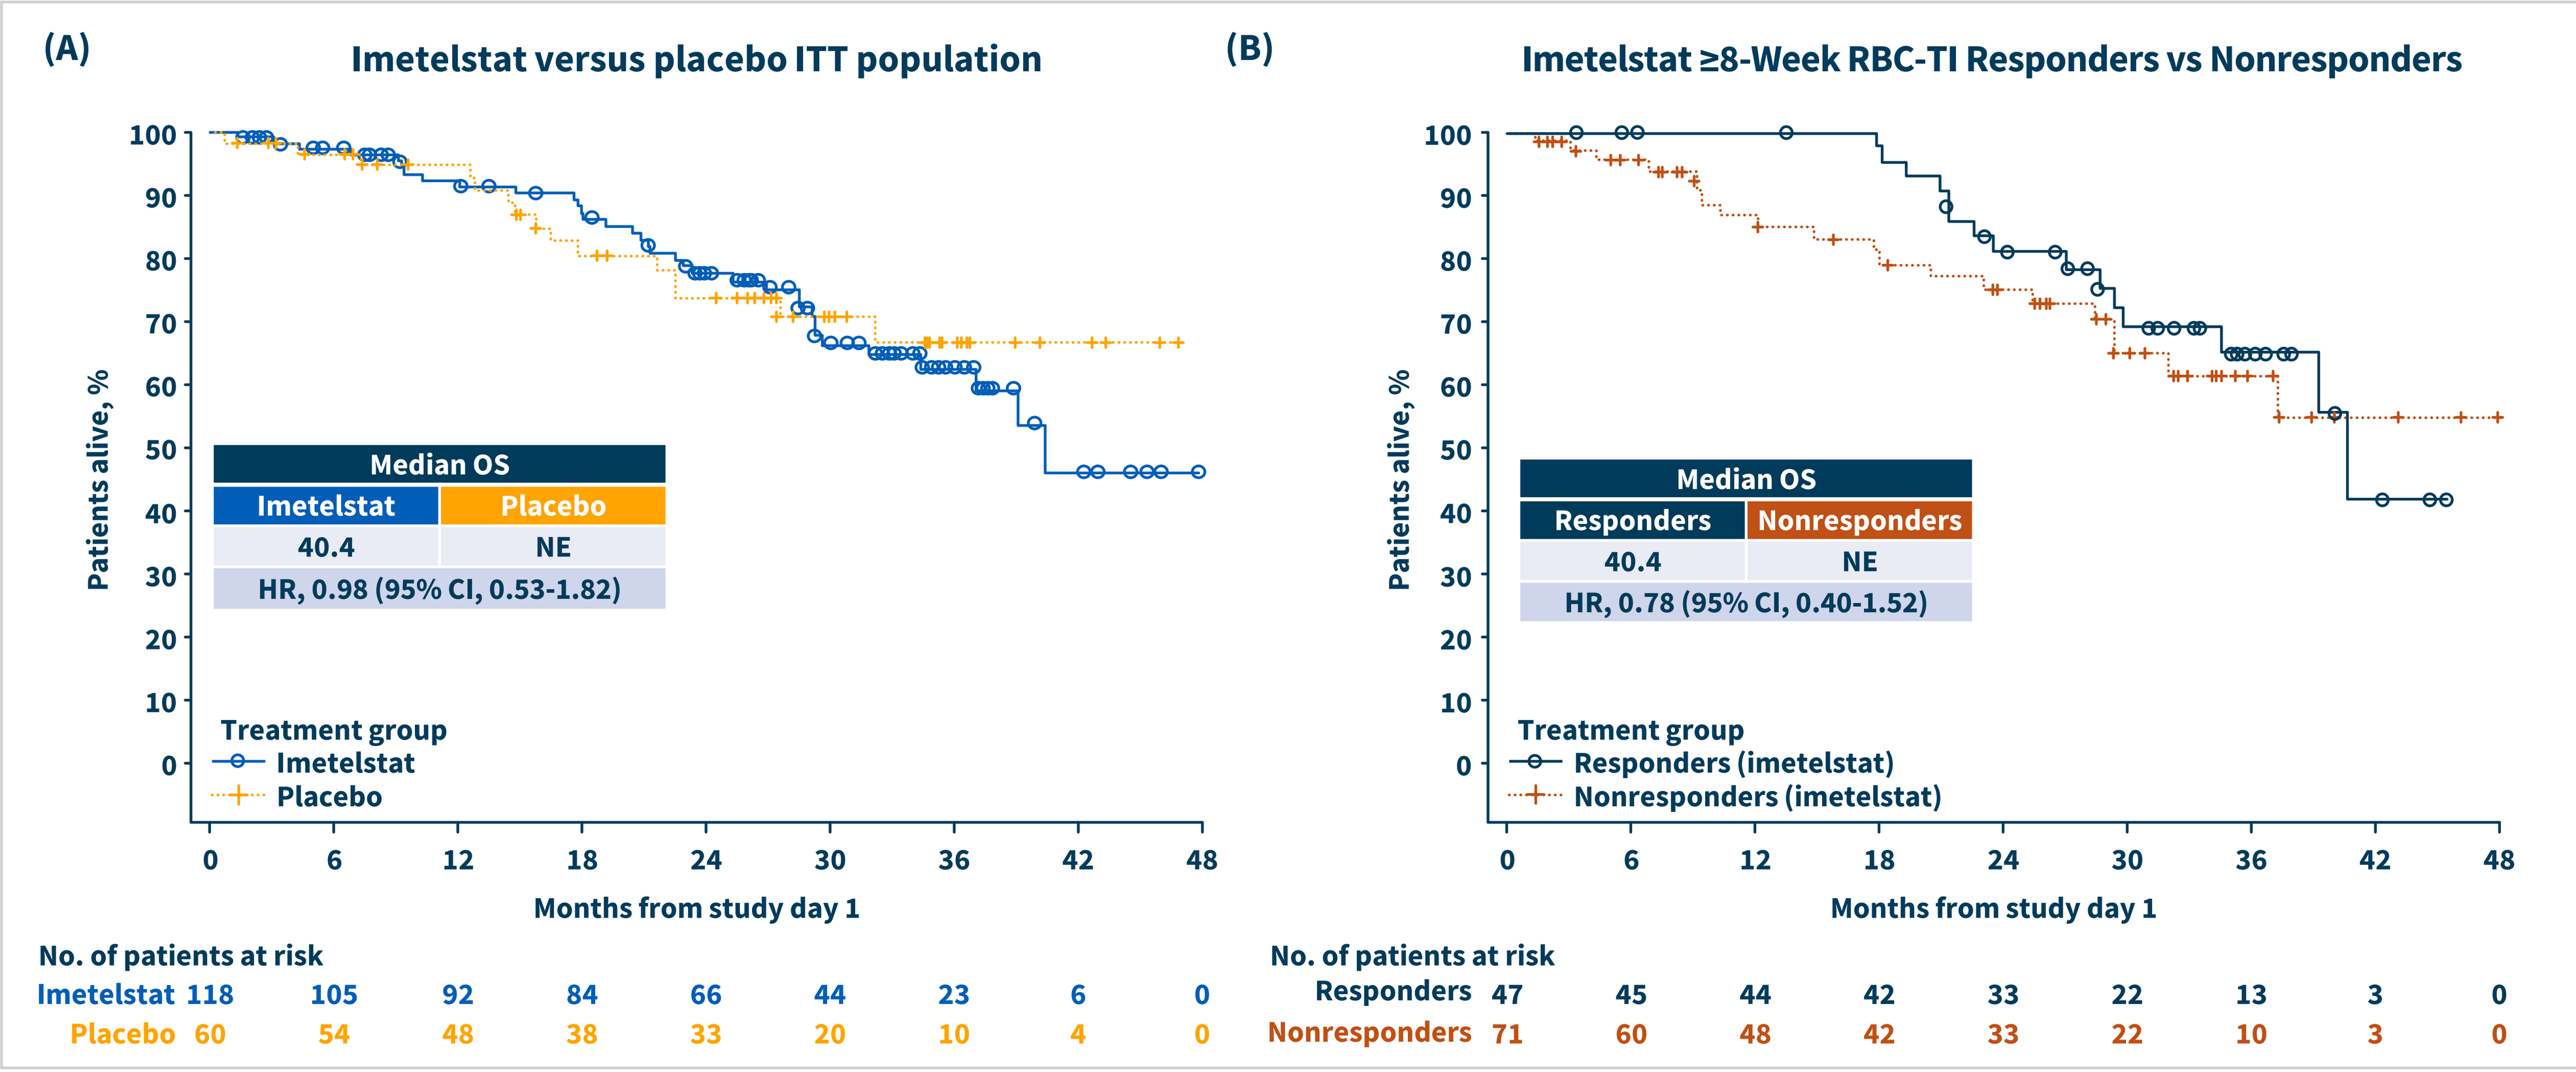
(A)
Imetelstat versus placebo ITT population
Patients alive, %
Median OS
Imetelstat Placebo
40.4 NE
HR, 0.98 (95% CI, 0.53-1.82)
Treatment group
Imetelstat
Placebo
0 6 12 18 24 30 36 42 48
Months from study day 1
No. of patients at risk
Imetelstat 118 105 92 84 66 44 23 6 0
Placebo 60 54 48 38 33 20 10 4 0
(B)
Imetelstat ≥8-Week RBC-TI Responders vs Nonresponders
Patients alive, %
Median OS
Responders Nonresponders
40.4 NE
HR, 0.78 (95% CI, 0.40-1.52)
Treatment group
Responders (imetelstat)
Nonresponders (imetelstat)
0 6 12 18 24 30 36 42 48
Months from study day 1
No. of patients at risk
Responders 47 45 44 42 33 22 13 3 0
Nonresponders 71 60 48 42 33 22 10 3 0

Exploratory analysis. Data cutoff date: January 2024.
HR, hazard ratio; ITT, intention-to-treat; NE, not estimable; OS, overall survival; RBC, red blood cell; TI, transfusion independence.

- Median estimated PFS has not been reached in either treatment arm (**Table 2**)
- The rate of progression to acute myeloid leukemia was low in both treatment arms (imetelstat [1.7%] and placebo [3.3%]) (**Table 2**)

Table 2. Disease Progression and Progression to AML

	Imetelstat (n=118)	Placebo (n=60)
PFS		
Number of PFS events, n (%)	29 (24.6)	14 (23.3)
Median (95% CI), months	NE (29.2-NE)	NE (16.7-NE)
HR (95% CI)[a] [*P* value[b]]	0.85 (0.44-1.64) [0.631]	
Disease progression, n (%)	13 (11.0)	8 (13.3)
Progression to AML		
n (%)	2 (1.7)	2 (3.3)
Median (95% CI), months	NE (NE-NE)	NE (NE-NE)
HR (95% CI)[a] [*P* value[b]]	0.45 (0.06-3.23) [0.418]	

Data cutoff date: January 2024.
[a]Cox proportional hazard model, stratified by prior RBC transfusion burden (≤6 U vs >6 U RBC) and International Prognostic Scoring System risk category (low vs intermediate-1), with treatment as the only covariate.
[b]Reported as descriptive *P* value.
AML, acute myeloid leukemia; HR, hazard ratio; NE, not estimable; PFS, progression-free survival.

Conclusions

- Results from these updated analyses confirm that achievement of RBC-TI with imetelstat was durable and associated with improvement in Hb level
 - Long-term RBC-TI responders achieved Hb levels that appeared to normalize
- In this LR-MDS population with high transfusion burden, rates of ≥8-week, ≥24-week, and ≥1-year RBC-TI concurrent with a rise in Hb of ≥1.5 g/dL were significantly[a] higher with imetelstat versus placebo
- OS analysis, with follow-up of 32 months, suggested no detriment for imetelstat-treated patients[a]
- These data further support the efficacy and clinical benefit of imetelstat in RBC-TD patients with LR-MDS R/R to or who were ineligible for ESAs

[a]≥1-year RBC-TI, Hb rise ≥1.5 g/dL, and OS are reported with descriptive statistics.

References

1. Platzbecker U, et al. *Leukemia*. 2021;35(8):2182-2198.
2. Harnan S, et al. *Acta Haematol*. 2016;136(1):23-42.
3. Asai A, et al. *Cancer Res*. 2003;63(14):3931-3939.
4. Herbert B-S, et al. *Oncogene*. 2005;24(33):5262-5268.
5. RYTELO (imetelstat). Package insert. Geron Corporation; 2024.
6. Platzbecker U, Santini V, et al. *Lancet*. 2024;403(10423):249-260.

Acknowledgments

- The authors thank all the patients and caregivers for their participation in this study and acknowledge the collaboration and commitment of all investigators and their research support staff
- All authors contributed to and approved the presentation; writing and editorial support were provided by Meredith Rogers, MS, CMPP, and Jeremy J. Henriques, PhD, CMPP, of The Lockwood Group (Stamford, CT, USA), funded by the Geron Corporation

Presented at the Society of Hematologic Oncology (SOHO) 2024 Annual Meeting; September 4-7, 2024; Houston, TX, USA & Online

ClinicalTrials.gov: https://clinicaltrials.gov/study/NCT02598661; email: medinfo@geron.com

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster

PosterSessionOnline.com
SOHO2024
Myelodysplastic Syndromes
Valeria Santini
MDS-157
SOHO2024