P05-21

Positive Results From REGENERATE: A Phase 3, International, Randomized, Placebo-Controlled Study Evaluating Obeticholic Acid Treatment for NASH

Zobair M. Younossi, Vlad Ratziu, Rohit Loomba, Mary Rinella, Quentin M. Anstee, Zachary Goodman, Pierre Bedossa, Andreas Geier, Susanne Beckebaum, Philip Newsome, David Sheridan, James Trotter, Whitfield Knapple, Eric Lawitz, Kris Kowdley, Aldo Montano-Loza, Jerome Boursier, Philippe Mathurin, Elisabetta Bugianesi, Giuseppe Mazzella, Antonio Olveira, Helena Cortez-Pinto, Isabel Graupera, David Orr, Lise Lotte Gluud, Jean-Francois Dufour, David Shapiro, Jason Campagna, Luna Zaru, Leigh MacConell, Reshma Shringarpure, Stephen Harrison, Arun J. Sanyal on behalf of the REGENERATE Study Investigators

Background and Rationale

REGENERATE STUDY OF OCA IN NASH

NASH: A Major Unmet Medical Need

- NASH is a growing and common cause of liver-related morbidity and mortality worldwide[1]
- NASH is projected to soon become the leading indication for liver transplantation in the United States[2]
- Fibrosis stage is the strongest predictor of adverse clinical outcomes in patients with NASH[3]
- There are currently no approved pharmacological therapies for NASH

Obeticholic Acid (OCA)

- OCA is a potent FXR agonist shown to improve NASH through multiple mechanisms in preclinical models, including a direct antifibrotic effect in the liver[4]
- In the Phase 2b FLINT study, 72 weeks of treatment with OCA 25 mg improved fibrosis and other histologic features of NASH[5]
- Based on the large unmet need and FLINT results, OCA has been designated a Breakthrough Therapy by the US FDA for the treatment of NASH patients with liver fibrosis

FXR, farnesoid X receptor.

REGENERATE STUDY DESIGN

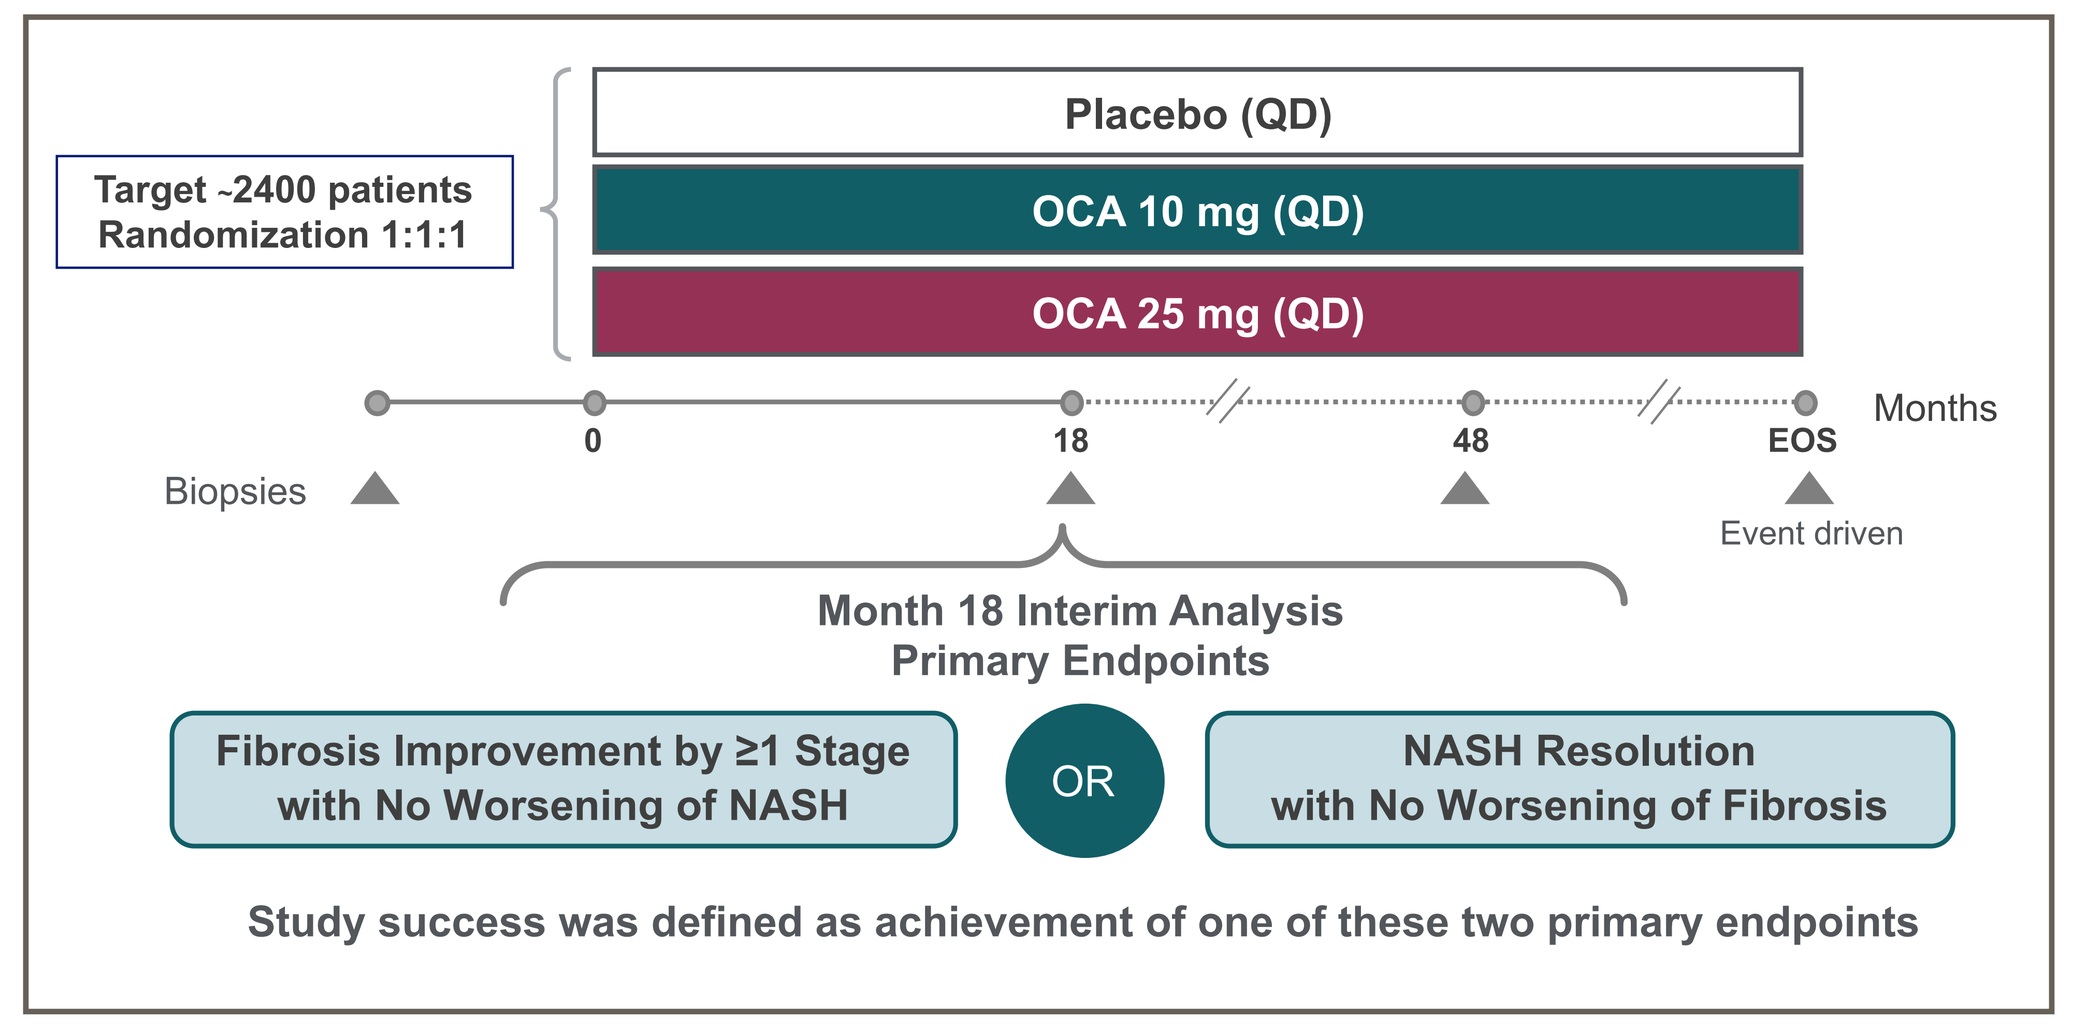
Target ~2400 patients
Randomization 1:1:1
Placebo (QD)
OCA 10 mg (QD)
OCA 25 mg (QD)
0
18
48
EOS
Months
Biopsies
Event driven
Month 18 Interim Analysis
Primary Endpoints
Fibrosis Improvement by ≥1 Stage with No Worsening of NASH
OR
NASH Resolution with No Worsening of Fibrosis
Study success was defined as achievement of one of these two primary endpoints

The interim analysis was conducted after 931 randomized patients with fibrosis stage 2 or 3 had or would have reached their actual/planned Month 18 visit (ITT population).
EOS analysis of clinical outcomes to confirm clinical benefit.
EOS, end of study; ITT, intent to treat; QD, once a day.

STUDY ELIGIBILITY CRITERIA

KEY INCLUSION CRITERIA

- Biopsy-confirmed NASH
- Fibrosis stage 2 or 3 (NASH CRN)
 - Exploratory cohort with fibrosis stage 1 and concomitant risk factors*
- NAFLD activity score (NAS) ≥4

KEY EXCLUSION CRITERIA

- Evidence of other chronic liver disease
- Histologic presence of cirrhosis
- Total bilirubin >1.5 mg/dL
- ALT ≥10 × ULN
- HbA1c >9.5%
- Significant alcohol consumption**

All biopsies were read centrally and at Month 18 biopsy slides were pair-read ensuring that pathologists were blinded to both treatment assignment and biopsy sequence

*Risk factors included type 2 diabetes, obesity (BMI ≥30 kg/m2) or ALT >1.5 × ULN.
**Defined as >2 units/day for females and >4 units/day for males for >3 months within 1 year before screening.
ALT, alanine aminotransferase; BMI, body mass index; CRN, clinical research network; HbA1c, glycated hemoglobin; NAFLD, nonalcoholic fatty liver disease; NAS, NAFLD activity score; ULN, upper limit of normal.

PATIENT DISPOSITION

ITT Population, N=931

Disposition, n (%)	Placebo (n=311)	OCA 10 mg (n=312)	OCA 25 mg (n=308)
Completed Month 18 biopsy	252 (81)	253 (81)	243 (79)
Study discontinuation	50 (16)	54 (17)	47 (15)
Treatment discontinuation	73 (23)	71 (23)	77 (25)
Withdrawal of consent	26 (8)	20 (6)	14 (5)
Adverse event	24 (8)	23 (7)	42 (14)
Physician decision	3 (<1)	1 (<1)	8 (3)
Lost to follow-up	7 (2)	7 (2)	5 (2)

Results

DEMOGRAPHIC AND BASELINE CHARACTERISTICS

ITT Population

Characteristics	Placebo (n=311)	OCA 10 mg (n=312)	OCA 25 mg (n=308)
Age, years, mean (SD)	55 (12)	55 (11)	55 (11)
Female, n (%)	187 (60)	177 (57)	175 (57)
White, n (%)	264 (94)	263 (92)	249 (87)
Hispanic ethnicity, n (%)	52 (18)	42 (15)	47 (17)
Fibrosis stage 3, n (%)	169 (54)	182 (58)	169 (55)
NAS ≥6, n (%)	215 (70)	211 (68)	208 (68)
Type 2 diabetes,* n (%)	175 (56)	171 (55)	171 (56)
Laboratory parameters, mean (SD)			
ALT, U/L	80 (57)	76 (47)	80 (56)
AST, U/L	59 (41)	57 (34)	57 (34)
Concomitant medication use			
Lipid lowering, n (%)	175 (56)	170 (54)	160 (52)
Statins, n (%)	144 (46)	142 (46)	127 (41)
Antidiabetic medication, n (%)	167 (54)	171 (55)	159 (52)
TZD,* n (%)	5 (2)	9 (3)	4 (1)
Vitamin E,* n (%)	42 (14)	34 (11)	32 (10)

ITT population, N=931.
*Randomization was stratified based on presence of type 2 diabetes and treatment with glitazones (TZDs) or Vitamin E.
AST, aspartate transaminase; SD, standard deviation; TZD, thiazolidinediones.

FIBROSIS IMPROVEMENT BY ≥1 STAGE WITH NO WORSENING OF NASH

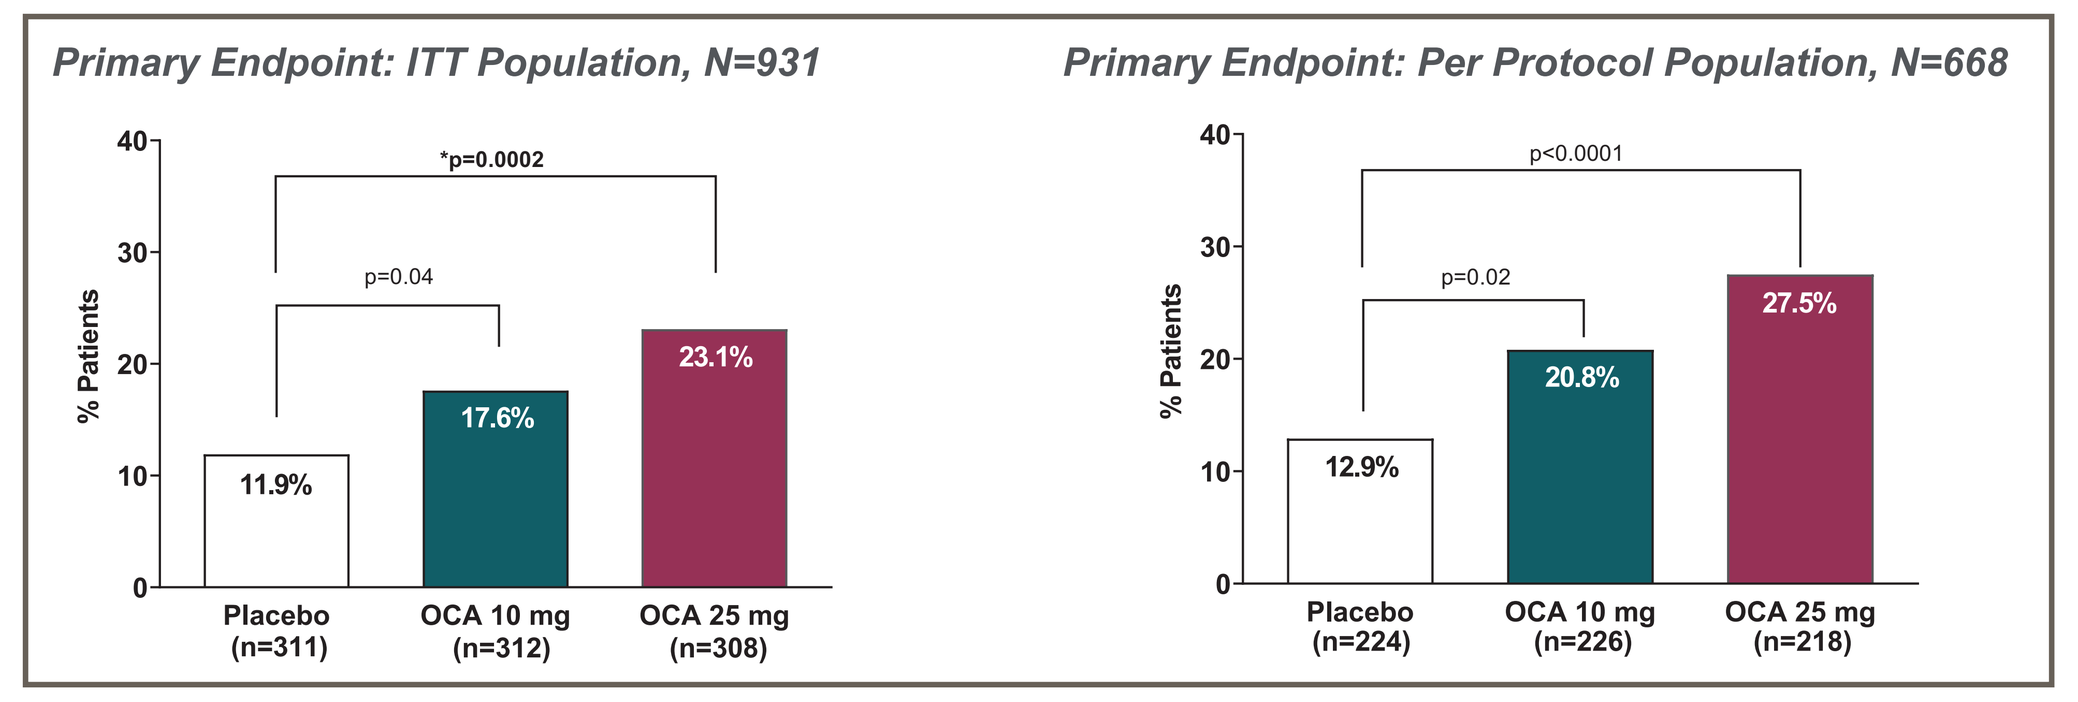
Primary Endpoint: ITT Population, N=931
% Patients
*p=0.0002
p=0.04
11.9%
17.6%
23.1%
Placebo (n=311)
OCA 10 mg (n=312)
OCA 25 mg (n=308)
Primary Endpoint: Per Protocol Population, N=668
p<0.0001
p=0.02
12.9%
20.8%
27.5%
Placebo (n=224)
OCA 10 mg (n=226)
OCA 25 mg (n=218)

Primary endpoint definition: fibrosis improvement by ≥1 stage (NASH CRN) with no worsening of NASH (defined as no worsening of hepatocellular ballooning, lobular inflammation or steatosis).
Study success was defined as achievement of one of the two primary endpoints evaluated in the Month 18 interim analysis.
*Statistically significant in accordance with the statistical analysis plan as agreed with the FDA. All other p values are nominal.
Per protocol population defined as all patients from the ITT population who completed ≥15 months of treatment and had a Month 18/end of treatment (EOT) biopsy, were on treatment for at least 30 days immediately preceding the biopsy, and did not have any major protocol deviation.
P values are nominal.

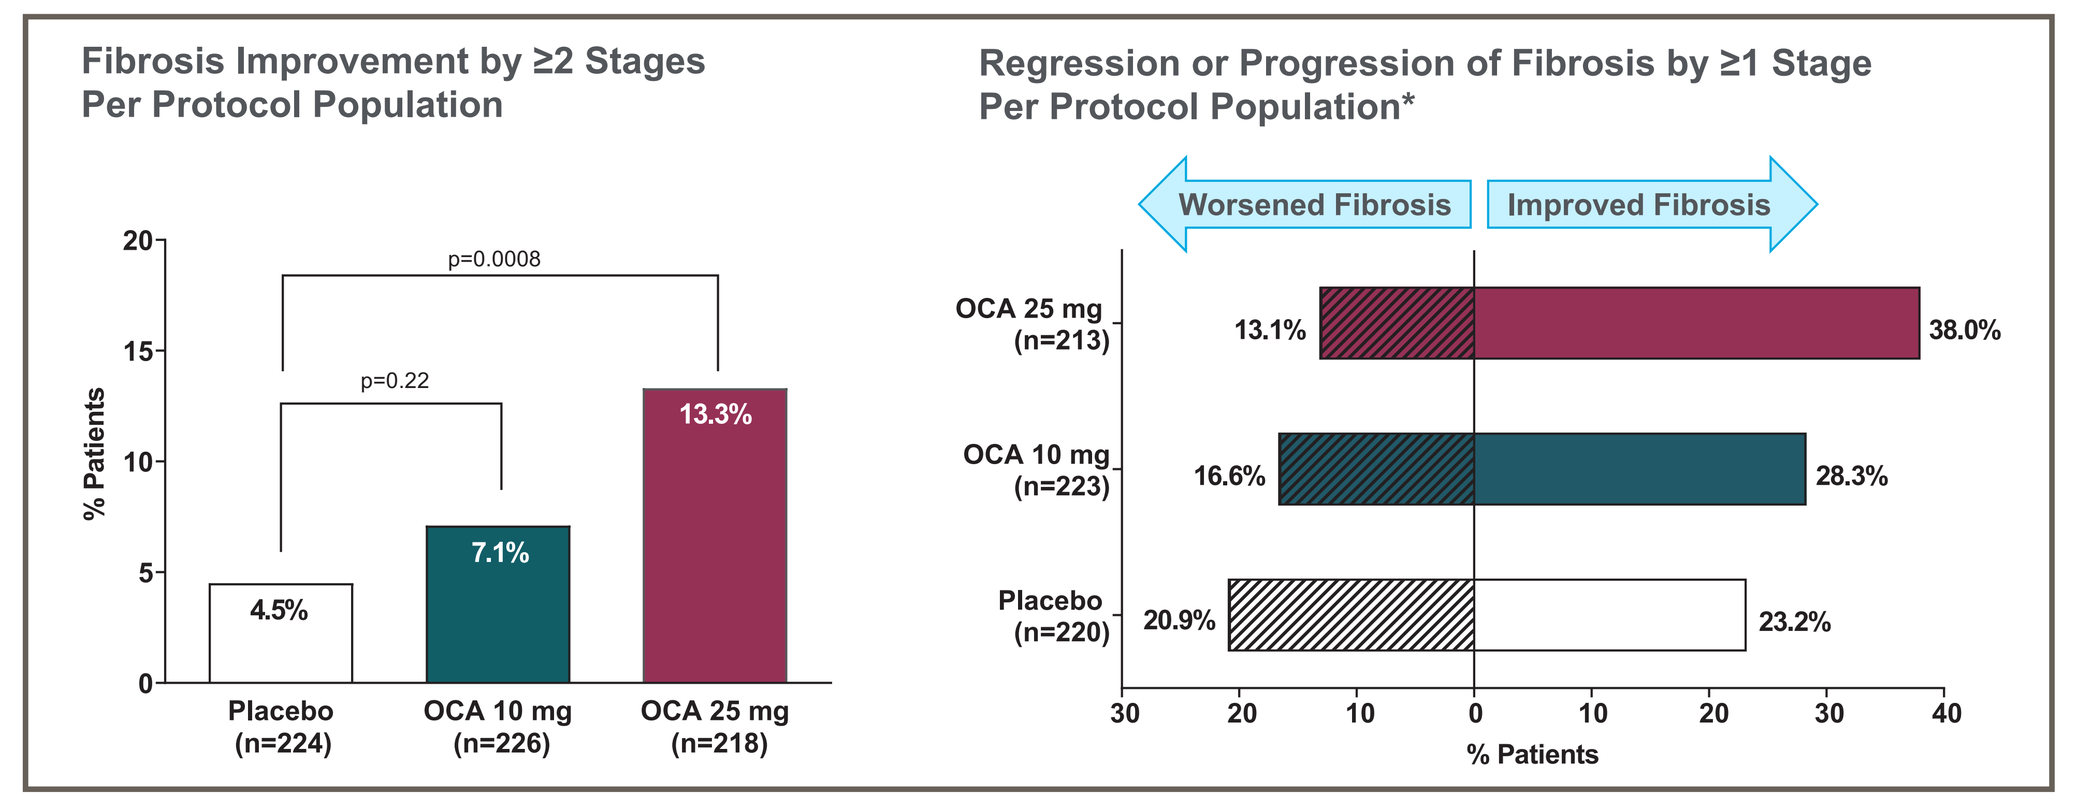
Fibrosis Improvement by ≥2 Stages
Per Protocol Population
% Patients
p=0.0008
p=0.22
4.5%
7.1%
13.3%
Placebo (n=224)
OCA 10 mg (n=226)
OCA 25 mg (n=218)
Regression or Progression of Fibrosis by ≥1 Stage
Per Protocol Population*
Worsened Fibrosis
Improved Fibrosis
OCA 25 mg (n=213)
OCA 10 mg (n=223)
Placebo (n=220)
13.1%
38.0%
16.6%
28.3%
20.9%
23.2%
% Patients

P values are nominal.
Per protocol population (N=668).
*Per protocol population with available fibrosis stage data at Month 18/EOT (n=656).

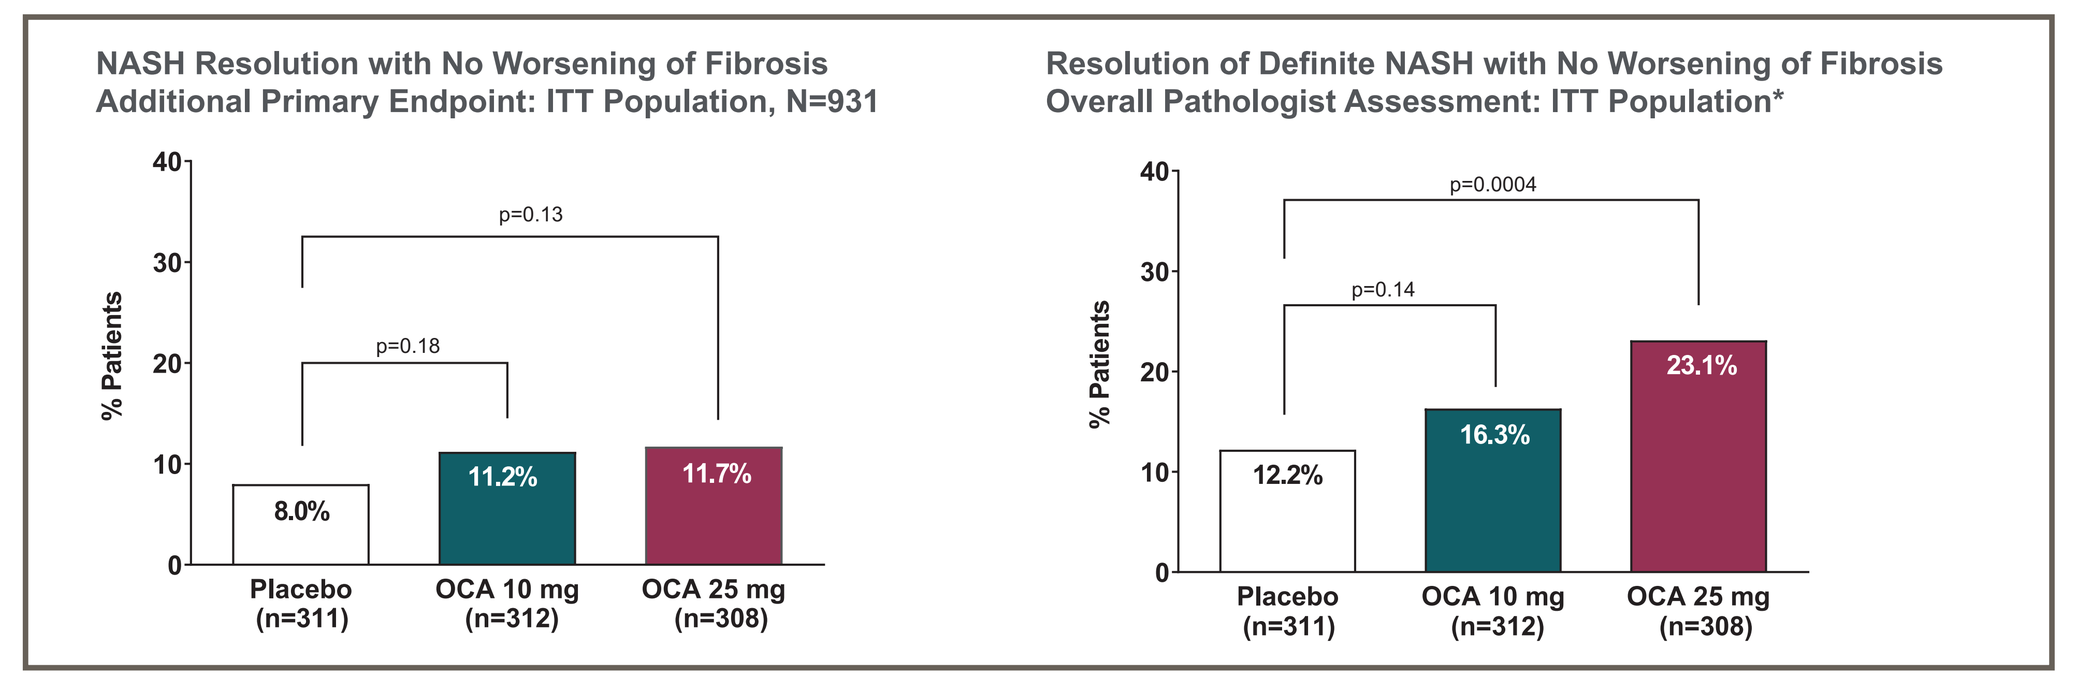
NASH Resolution with No Worsening of Fibrosis
Additional Primary Endpoint: ITT Population, N=931
% Patients
p=0.13
p=0.18
8.0%
11.2%
11.7%
Placebo (n=311)
OCA 10 mg (n=312)
OCA 25 mg (n=308)
Resolution of Definite NASH with No Worsening of Fibrosis
Overall Pathologist Assessment: ITT Population*
p=0.0004
p=0.14
12.2%
16.3%
23.1%

Primary endpoint definition: (i) overall pathologist assessment of "no steatohepatitis," and (ii) hepatocellular ballooning = 0 and lobular inflammation = 0 or 1, and (iii) no increase in fibrosis stage from baseline.
Study success was defined as achievement of one of the two primary endpoints evaluated in the Month 18 interim analysis.
*Post-hoc analysis with endpoint defined as: (i) overall pathologist assessment of "no steatohepatitis," and (ii) no increase in fibrosis stage from baseline.
P values are nominal.

CHANGES IN LIVER BIOCHEMISTRY OVER TIME

Per Protocol Population

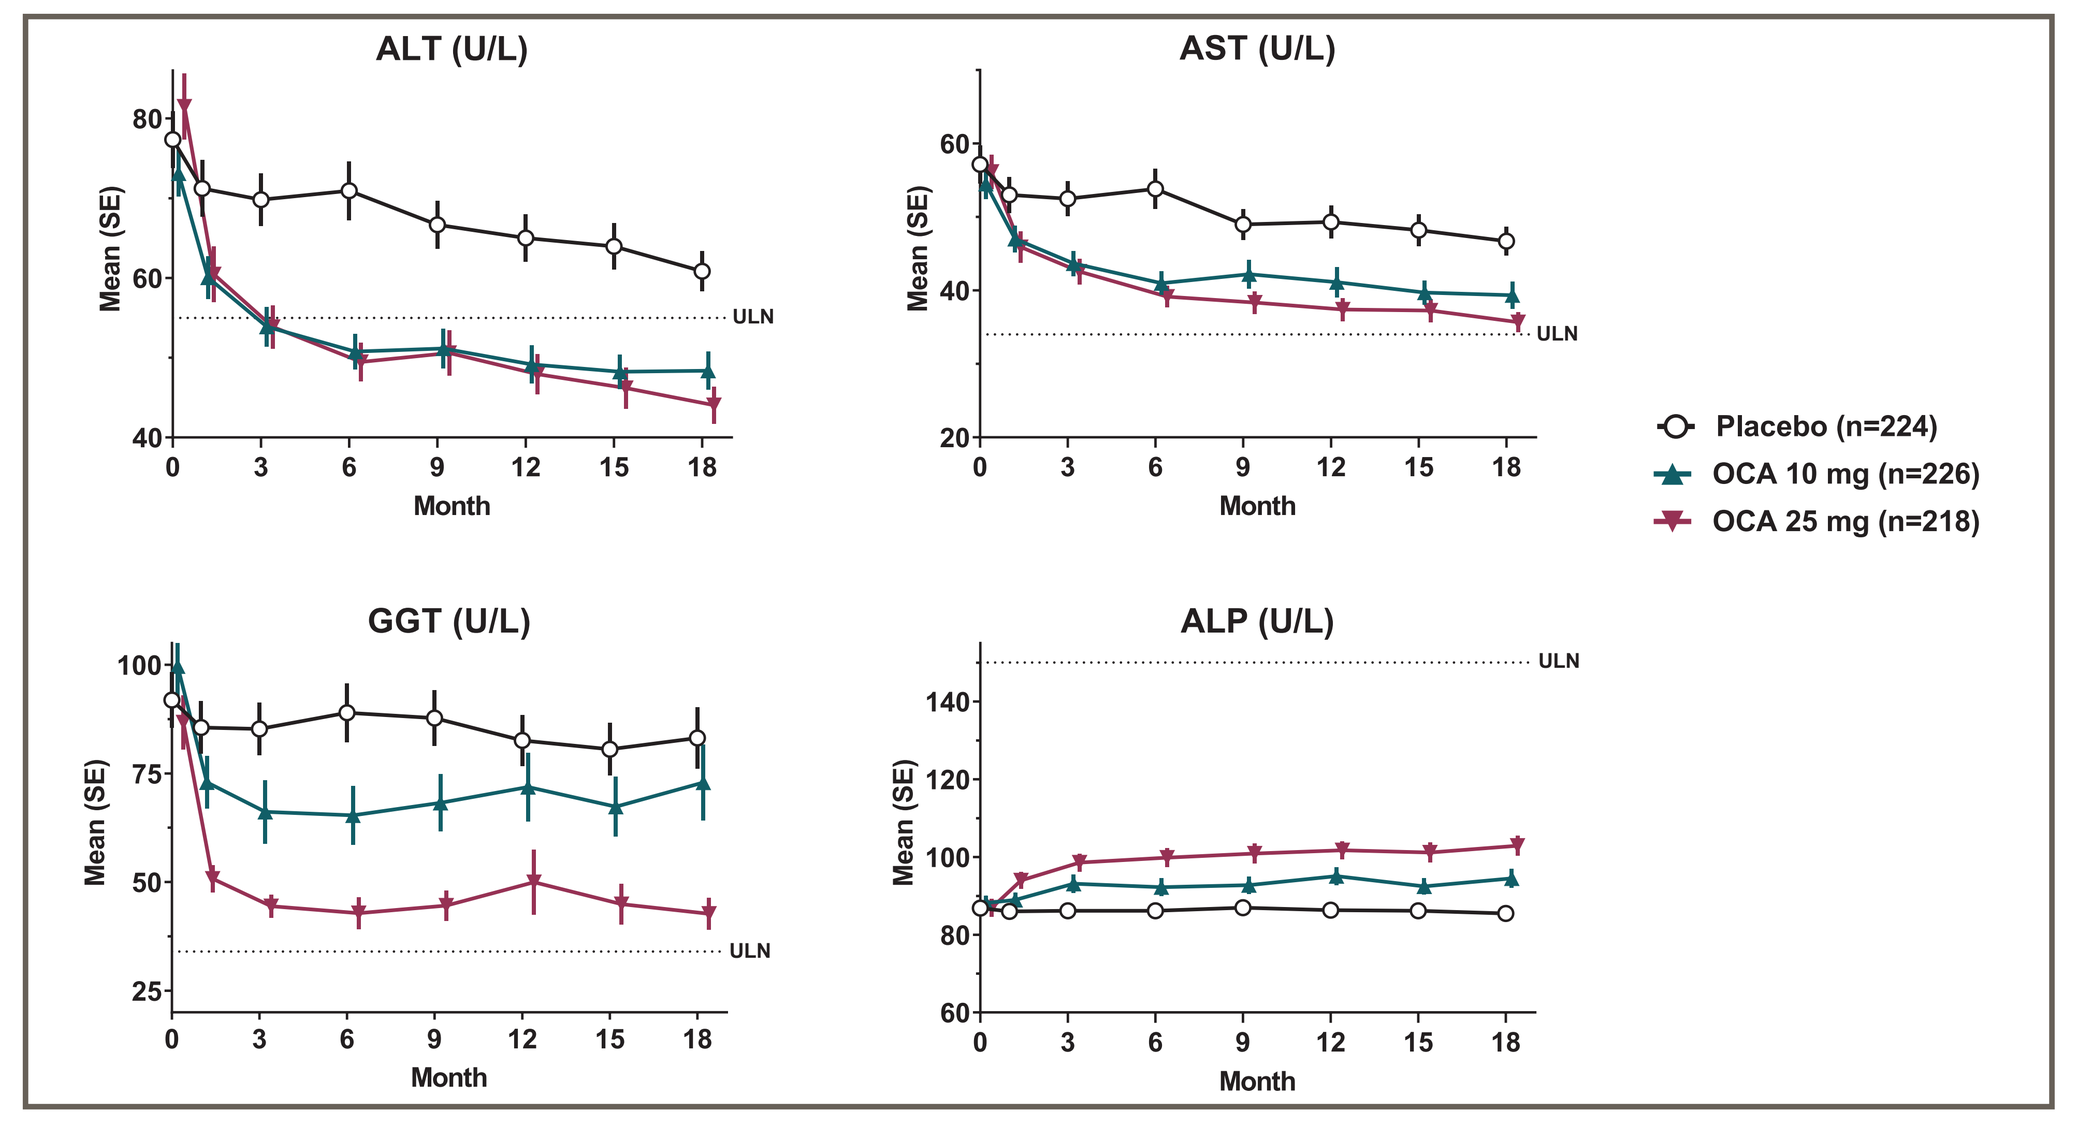
ALT (U/L)
AST (U/L)
GGT (U/L)
ALP (U/L)
Mean (SE)
Month
ULN
Placebo (n=224)
OCA 10 mg (n=226)
OCA 25 mg (n=218)

Per protocol population (N=668). ALP, alkaline phosphatase; GGT, gamma-glutamyl transferase.

SUMMARY OF TREATMENT-EMERGENT ADVERSE EVENTS

Safety Population

n (%)	Placebo (n=657)	OCA 10 mg (n=653)	OCA 25 mg (n=658)
≥1 Treatment-Emergent Adverse Event (TEAE)	548 (83)	579 (89)	601 (91)
TEAEs by Severity			
Mild	160 (24)	163 (25)	130 (20)
Moderate	294 (45)	323 (49)	338 (51)
Severe	87 (13)	89 (14)	130 (20)
TEAEs Leading to Treatment Discontinuation	41 (6)	39 (6)	83 (13)
Serious Adverse Events (SAEs)	75 (11)	72 (11)	93 (14)
Deaths	2 (<1)	0	1 (<1)

AEs were mostly mild to moderate in severity
The frequency of SAEs was similar across treatment arms
No single SAE occurred in >1% of patients in any treatment arm

Safety population (N=1968).
AE, adverse event.

MOST FREQUENT TREATMENT-EMERGENT ADVERSE EVENTS

Safety Population: Events Occurring in ≥10% of Patients in Any Treatment Group

n (%)	Placebo (n=657)	OCA 10 mg (n=653)	OCA 25 mg (n=658)
Pruritus (all pooled terms)	123 (19)	183 (28)	336 (51)
LDL increased	47 (7)	109 (17)	115 (17)
Nausea	77 (12)	72 (11)	83 (13)
Fatigue	88 (13)	78 (12)	71 (11)
Constipation	36 (5)	65 (10)	70 (11)
Abdominal pain	62 (9)	66 (10)	67 (10)
Diarrhea	79 (12)	44 (7)	49 (7)

Most frequent TEAEs were mild to moderate in severity and consistent with the known profile of OCA

Data are presented in decreasing order of occurrence in the OCA 25 mg group. All data are based on investigator-reported events.
Safety population (N=1968).
LDL, low density lipoprotein.

Additional Safety and Tolerability Information

SAFETY POPULATION

Pruritus

- Incidence was highest in the first 3 months and decreased thereafter
- In patients on OCA 25 mg reporting pruritus, 93% of events were mild to moderate
- 9% of patients on OCA 25 mg discontinued due to pruritus: more than half of these were protocol mandated and overall discontinuation rates were similar across the treatment arms

Hepatobiliary

- Hepatic TEAEs were balanced across treatment groups (Placebo, 13%; OCA 10 mg, 13%; OCA 25 mg, 11%)
- Hepatic SAEs were rare (<1% in all treatment groups): more occurred in the OCA 25 mg group with no pattern attributable to OCA (based on eDISH and case review)
- Incidence of cholelithiasis or cholecystitis AEs* was low (Placebo, <1%; OCA 10 mg, 1%; OCA 25 mg, 3%)

Cardiovascular

- Incidence of CV** SAEs was low and balanced across groups (Placebo, 2%; OCA 10 mg, 1%; OCA 25 mg, 2%)

*Gallbladder SMQ includes TEAEs and SAEs.
**By SMQ and preferred term.
CV, cardiovascular; eDISH, Evaluation of Drug-Induced Serious Hepatotoxicity; SMQ, standardized MedDRA queries.

Summary and Conclusion

REGENERATE IS THE FIRST SUCCESSFUL PHASE 3 STUDY IN PATIENTS WITH NASH

- OCA 25 mg met the primary fibrosis endpoint at the Month 18 interim analysis
- The antifibrotic effect was dose dependent and consistent across endpoints and key subgroups
- Although the primary NASH resolution endpoint was not met, OCA ameliorated steatohepatitis based on pathologist overall assessment and improvement in key disease activity parameters
- OCA rapidly and sustainably improved ALT, AST and GGT
- AEs were mostly mild to moderate; the most common were consistent with the known profile of OCA
- The study is ongoing to confirm benefit on clinically important outcomes

References

1. Angulo P, et al. *Gastroenterology*. 2015;149:389-397.
2. Diehl AM, et al. *N Engl J Med*. 2017;377:2063-2072.
3. Dulai PS, et al. *Hepatology*. 2017;65(5):1557–1565.
4. Albanis A, et al. *Hepatology*. 2005;42:1040A.
5. Neuschwander-Tetri BA, et al. *Lancet*. 2015;385:956-965.

Disclosures

ZMY – Consults for BMS, Echosens, Gilead, GSK, Intercept, Merck, Novartis, Novo Nordisk, Quest Diagnostics, Siemens, Shionogi, Terns and Viking; Receives Grant/Research Support from BMS, Gilead and Intercept

Corresponding Author

Zobair M. Younossi (zobair.younossi@inova.org)

Clinical Trial Information: CT.gov: NCT02548351; Eudra CT: 2015-002560-16

Intercept

Presented at EASL NAFLD Summit • September 26–28, 2019 • Seville, Spain

PosterSessionOnline.com
Poster presented at: EASL The Home of Hepatology | SUMMIT
Metabolism, alcohol and toxicity
Aldo Trylesinski
P05-21
NAFLD2019