The Value of sPESI for Risk Stratification in Patients with Pulmonary Embolism

Phil Wells, MD, FRCPC MSc[1]; W. Frank Peacock, MD, FACEP[2]; Gregory J. Fermann, MD, FACEP[3]; Craig I. Coleman, PharmD, FASHP[4]; Li Wang, MA, PhD, MBA[5]; Onur Baser, MA, MS, PhD[6,7]; Jeff Schein, DrPH, MPH[8]; Concetta Crivera, PharmD, MPH[8]

[1]University of Ottawa and the Ottawa Hospital Research Institute, Ottawa, ON, Canada; [2]Baylor College of Medicine, Houston, TX, USA; [3]University of Cincinnati, OH, USA; [4]University of Connecticut, Hartford, USA; [5]STATinMED Research, Analytic Research, Plano, TX, USA; [6]Columbia University, Innovation & Outcomes Research, Department of Surgery, New York, NY, USA; [7]STATinMED Research, Health Economics & Outcomes Research, New York, NY, USA; [8]Janssen Scientific Affairs, LLC, Raritan, NJ, USA

BACKGROUND

- Pulmonary embolism (PE) is responsible for 300,000 US deaths each year. Initial PE-related hospitalization costs have been estimated at $13,300-$31,000 annually.
- The key to effective treatment lies in appropriate risk stratification, where low-risk PE (LRPE) patients can benefit from abbreviated hospital stays or outpatient therapy, which could substantially reduce the clinical and economic disease burden.
- Several risk-stratification algorithms have been developed, including the Geneva score, the Pulmonary Embolism Severity Index (PESI) score, the simplified PESI (sPESI) score, the Spanish score, the Davies criteria, the Home management exclusion criteria, and the Hestia criteria.
- Current data is limited regarding the use of the sPESI score for risk stratification.

OBJECTIVES

Assess PE patients in the Veterans Health Administration (VHA) population using the sPESI score as a risk-stratification method.

METHODS

Data Source

- VHA Medical SAS Administrative Database from 01OCT2010-30SEP2015

Sample Selection

Figure 1. Patient Selection Criteria

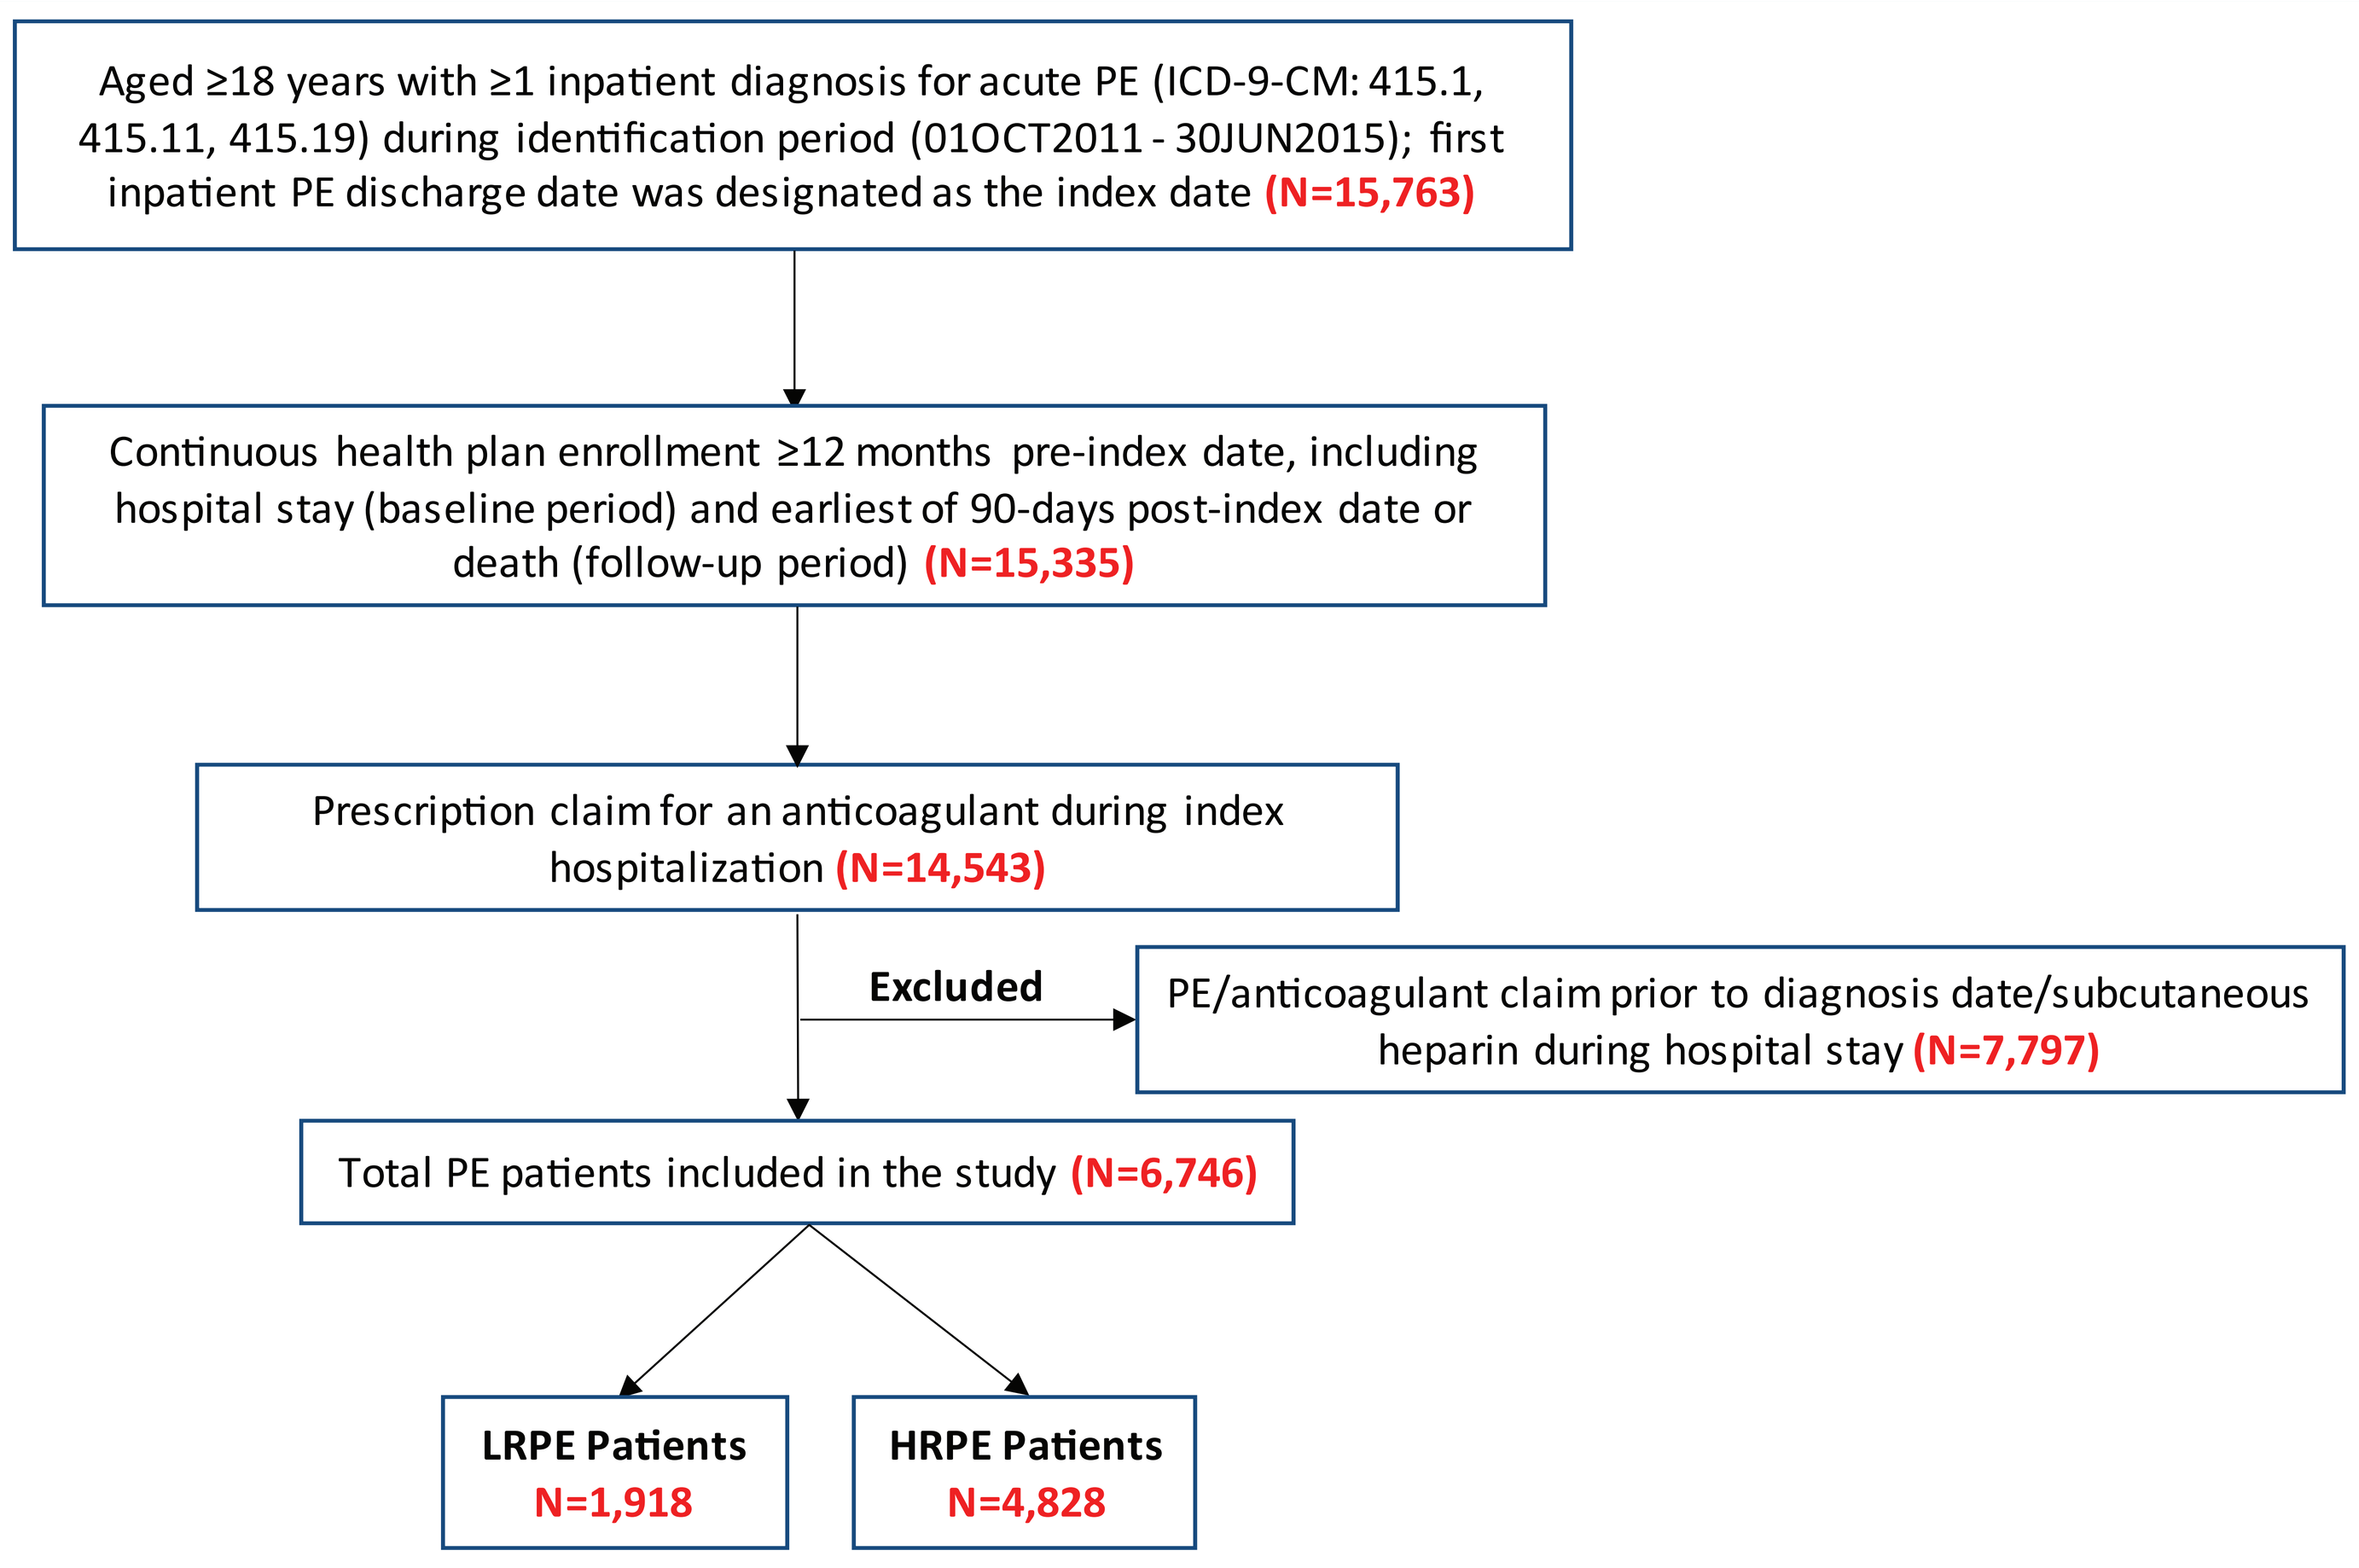
Aged ≥18 years with ≥1 inpatient diagnosis for acute PE (ICD-9-CM: 415.1, 415.11, 415.19) during identification period (01OCT2011 - 30JUN2015); first inpatient PE discharge date was designated as the index date (N=15,763)
Continuous health plan enrollment ≥12 months pre-index date, including hospital stay (baseline period) and earliest of 90-days post-index date or death (follow-up period) (N=15,335)
Prescription claim for an anticoagulant during index hospitalization (N=14,543)
Excluded
PE/anticoagulant claim prior to diagnosis date/subcutaneous heparin during hospital stay (N=7,797)
Total PE patients included in the study (N=6,746)
LRPE Patients N=1,918
HRPE Patients N=4,828

HRPE: high-risk pulmonary embolism; LRPE: low-risk pulmonary embolism; PE: pulmonary embolism

Cohort Assignment

- Patients were stratified into LRPE and high-risk PE (HRPE) cohorts using the sPESI criteria. sPESI is a simplified version of the PESI in which selected variables of the original score are included (age, history of cancer, history of chronic cardiopulmonary disease, pulse, systolic blood pressure, oxygen saturation levels).
- PE patients scoring 0 points were considered low risk; all others were considered high risk.

Outcome Measures

- Hospital-acquired complications (HACs) during index hospitalization included any of the following, using pre-specified ICD-9-CM codes: catheter-associated urinary tract infection, methicillin-resistant *Staphylococcus aureus*, *Clostridium difficile* infection, hospital-acquired bacterial pneumonia, foreign object retained after surgery, air embolism, blood incompatibility, pressure ulcer (stages III & IV), trauma/injury, procedure-related, poor glycemic control, iatrogenic pneumothorax with venous catheterization, vascular catheter-associated infection, or surgical site infection. Any condition with no patients was omitted.
- PE-related outcomes (recurrent venous thromboembolism [VTE], major bleeding, or death) as well as all-cause health care resource utilization and costs (inpatient stays, outpatient visits, and other services) during the 90-day follow-up period.

Statistical Analysis

- Descriptive statistics were provided for all study variables.
- Chi-square tests were for categorical variables.
- Student t-tests were used for continuous variables.

RESULTS

Total: 6,746 PE patients

- 4,828 HRPE patients
- 1,918 LRPE patients (Figure 1)

Baseline Demographic and Clinical Characteristics

In this VHA sample, most patients were male (6434; 95.38%) and either white (4565; 67.67%) or black (1486; 22.03%). HRPE patients were more likely to have higher Charlson comorbidity scores and suffer from myocardial infarction, congestive heart failure, peripheral vascular disease, cerebrovascular disease, chronic obstructive pulmonary disease, moderate/severe renal disease, diabetes, and cancer (all p<0.001). HRPE and LRPE patients had similar rates of dementia, rheumatic disease, peptic ulcer disease, moderate-severe liver disease, and AIDS.

HACs, 90-day Follow-up Outcomes

During the index hospitalization, LRPE patients had:

- fewer HACs;
- fewer bacterial pneumonia diagnoses; and
- shorter inpatient length of stay (Table 1).

RESULTS (cont'd)

During the 90-day follow-up period, LRPE patients had:

- fewer PE-related outcomes, including recurrent VTE, major bleeding, or death (Table 1);
- fewer inpatient, outpatient, and pharmacy visits per patient (Figure 2); and
- lower health care costs (Figure 3).

Table 1. Hospital-acquired Complications and PE-related Outcomes among LRPE vs HRPE Patients

	LRPE Cohort N=(1918)		HRPE Cohort N=(4828)			
	N/Mean	%/STD	N/Mean	%/STD	p-value	SD
Outcomes during the index hospitalization						
Hospital acquired complications, ANY	218	11.37%	964	19.97%	<0.0001	23.83
Catheter-associated Urinary Tract Infection	4	0.21%	12	0.25%	0.7606	0.84
Methicillin-resistant Staphylococcus Aureus (MRSA)	22	1.15%	54	1.12%	0.9202	0.27
Clostridium Difficile Infection	14	0.73%	65	1.35%	0.0338	6.08
Hospital Acquired (Bacterial) Pneumonia	128	6.67%	711	14.73%	<0.0001	26.27
Foreign Object Retained After Surgery	1	0.05%	1	0.02%	0.4989	1.65
Pressure Ulcer Stages III & IV	7	0.36%	25	0.52%	0.4098	2.31
Trauma/Injury	57	2.97%	155	3.21%	0.6124	1.38
Poor Glycemic Control	7	0.36%	28	0.58%	0.2676	3.14
Latrogenic Pneumothorax with Venous Catheterization	1	0.05%	19	0.39%	0.0200	7.24
Vascular Catheter-associated Infection	5	0.26%	10	0.21%	0.6735	1.11
Surgical Site Infection	3	0.16%	4	0.08%	0.3973	2.13
Bacterial Pneumonia	203	10.58%	1078	22.33%	<0.0001	32.07
Inpatient length of stay (in days)	1.92	8.30	2.54	8.44	0.0064	7.38
PE-related Clinical Outcomes during the 90-day follow-up period						
Recurrent VTE	84	4.38%	291	6.03%	0.0077	7.42
Time to first VTE, days	32.58	24.49	32.28	21.69	0.9133	1.30
Major Bleeding	23	1.20%	93	1.93%	0.0382	5.86
Time to first Major Bleeding, days	25.78	22.09	29.15	26.01	0.5687	13.96
Death	71	3.70%	780	16.16%	<0.0001	42.57
Time to Death, days	32.10	23.01	32.25	22.14	0.9554	0.68

SD: standard deviation; STD: standardized difference; VTE: venous thromboembolism

Figure 2. Health Care Utilization among LRPE vs HRPE Patients during the 90-day Follow-up Period

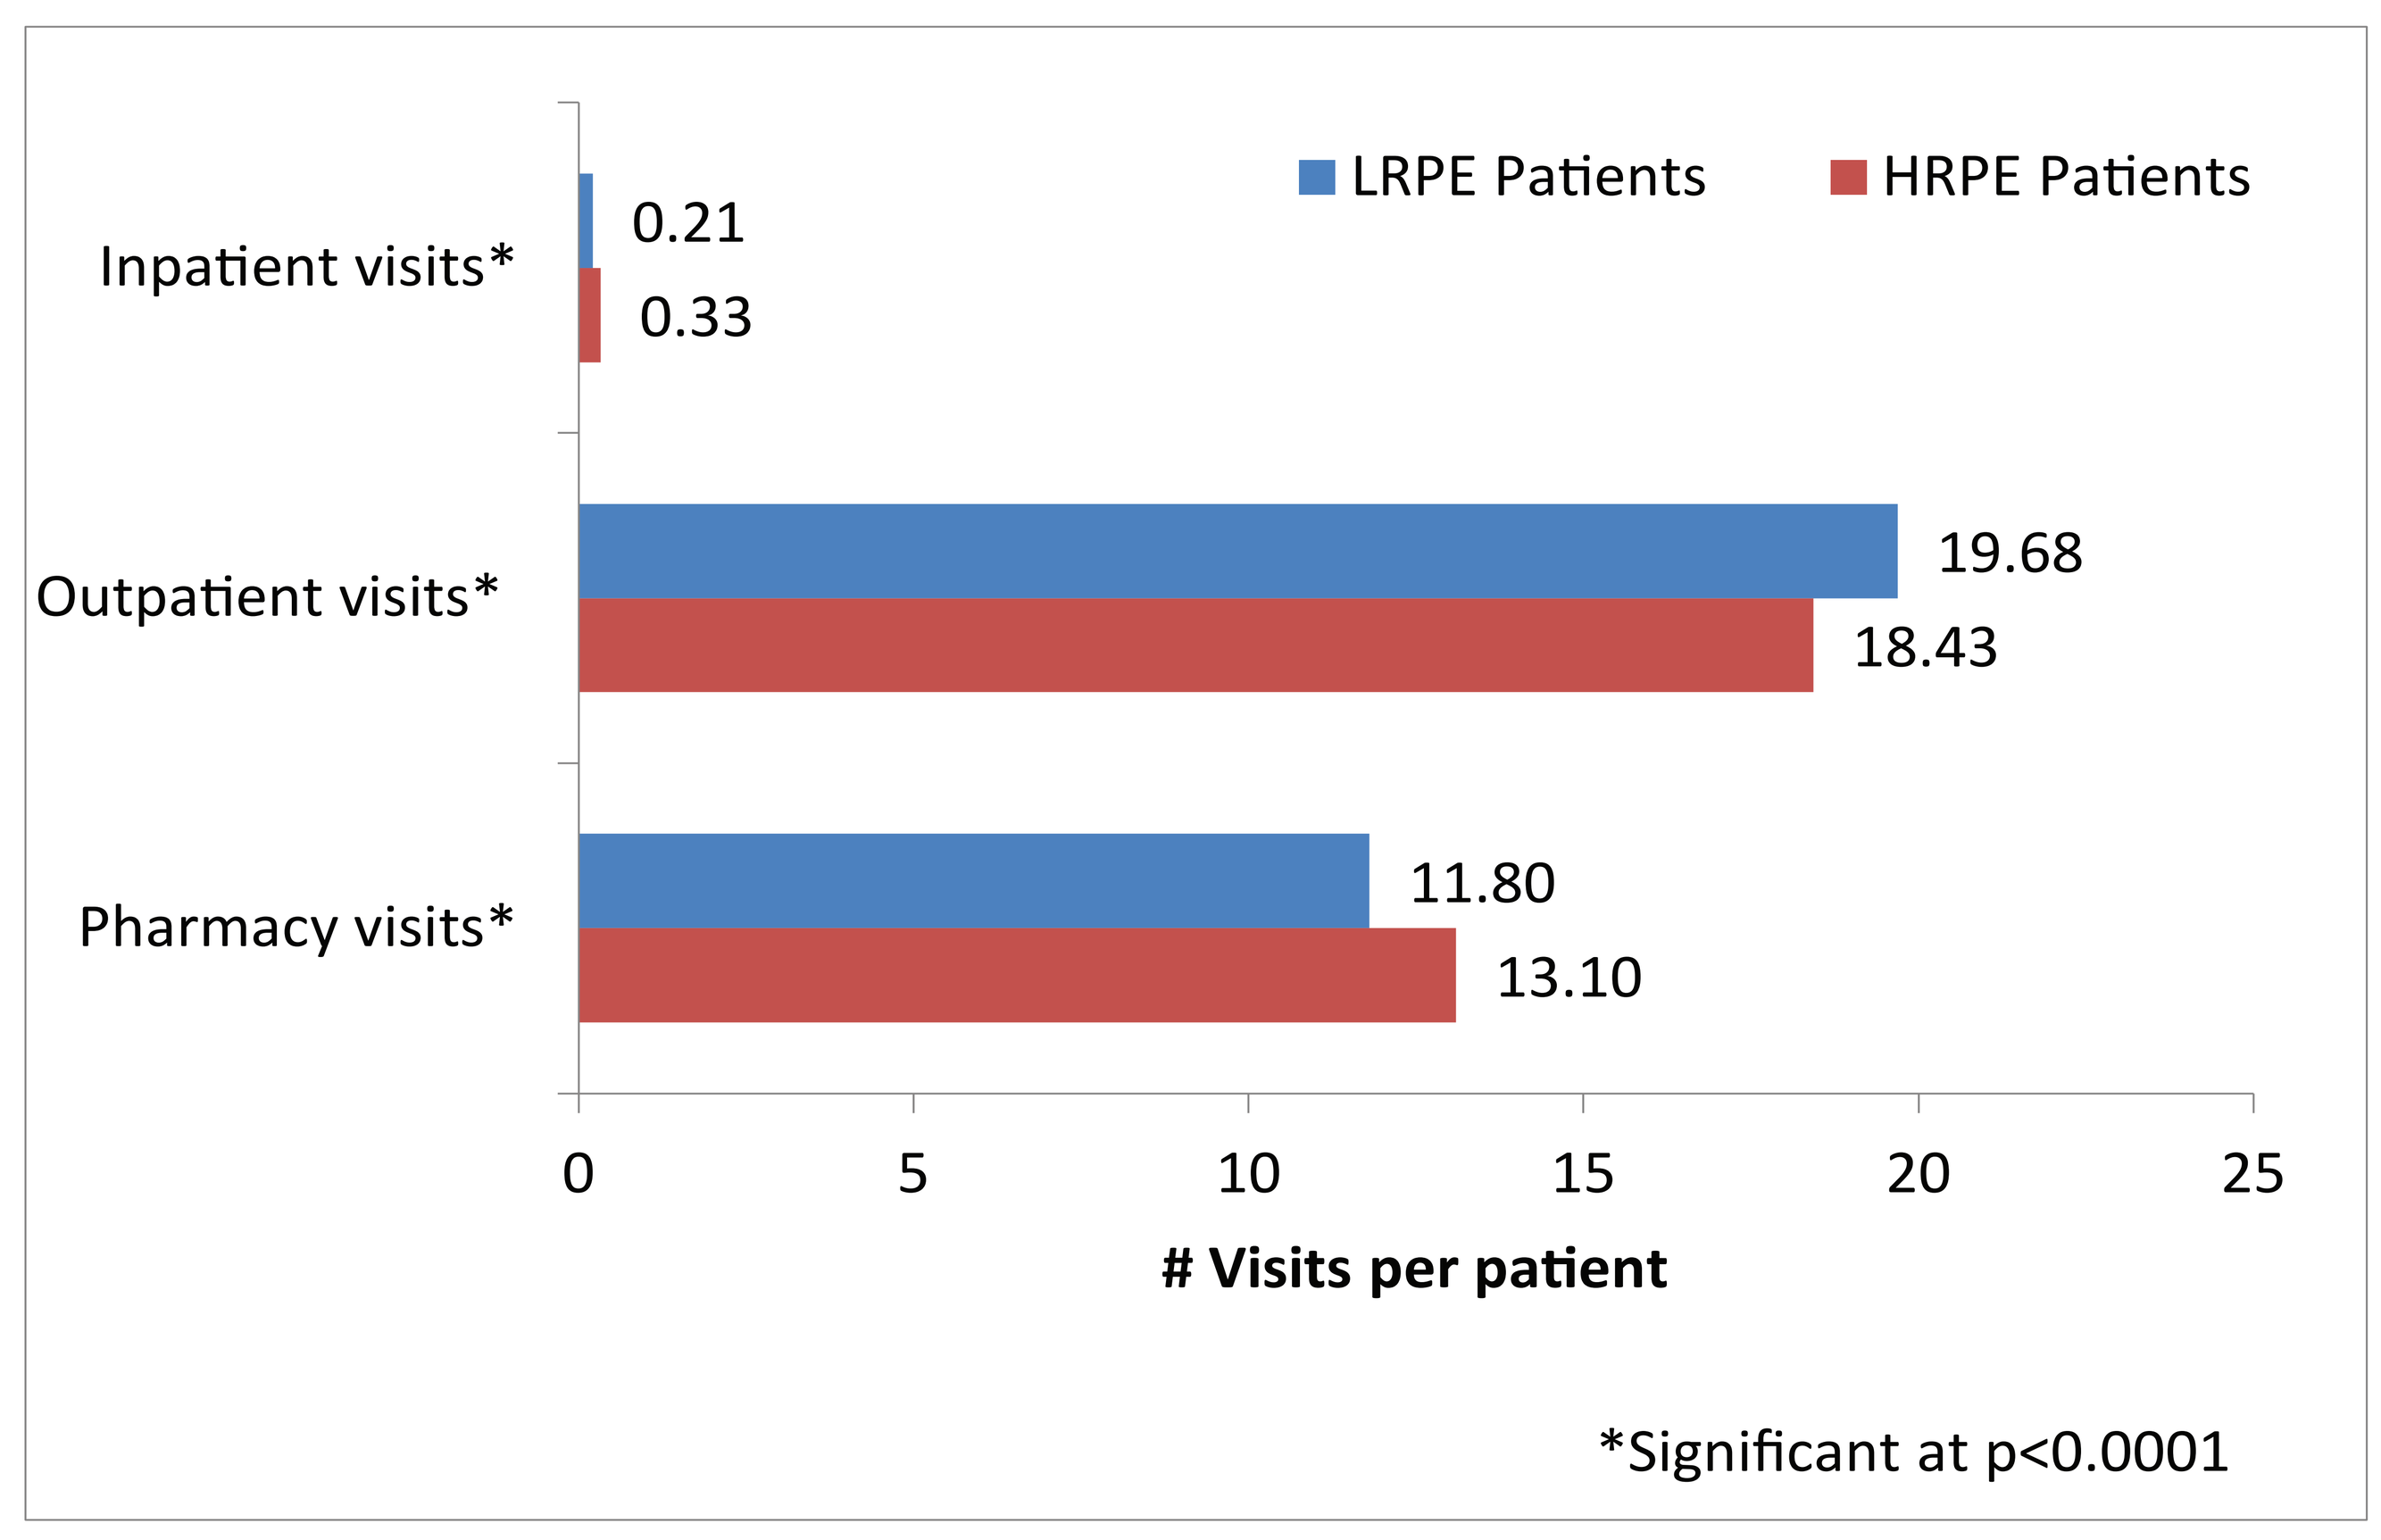
LRPE Patients
HRPE Patients
Inpatient visits*
0.21
0.33
Outpatient visits*
19.68
18.43
Pharmacy visits*
11.80
13.10
0
5
10
15
20
25
Visits per patient
*Significant at p<0.0001

HRPE: high-risk pulmonary embolism; LRPE: low-risk pulmonary embolism

Figure 3. Health Care Costs among LRPE vs HRPE Patients during the 90-day Follow-up Period

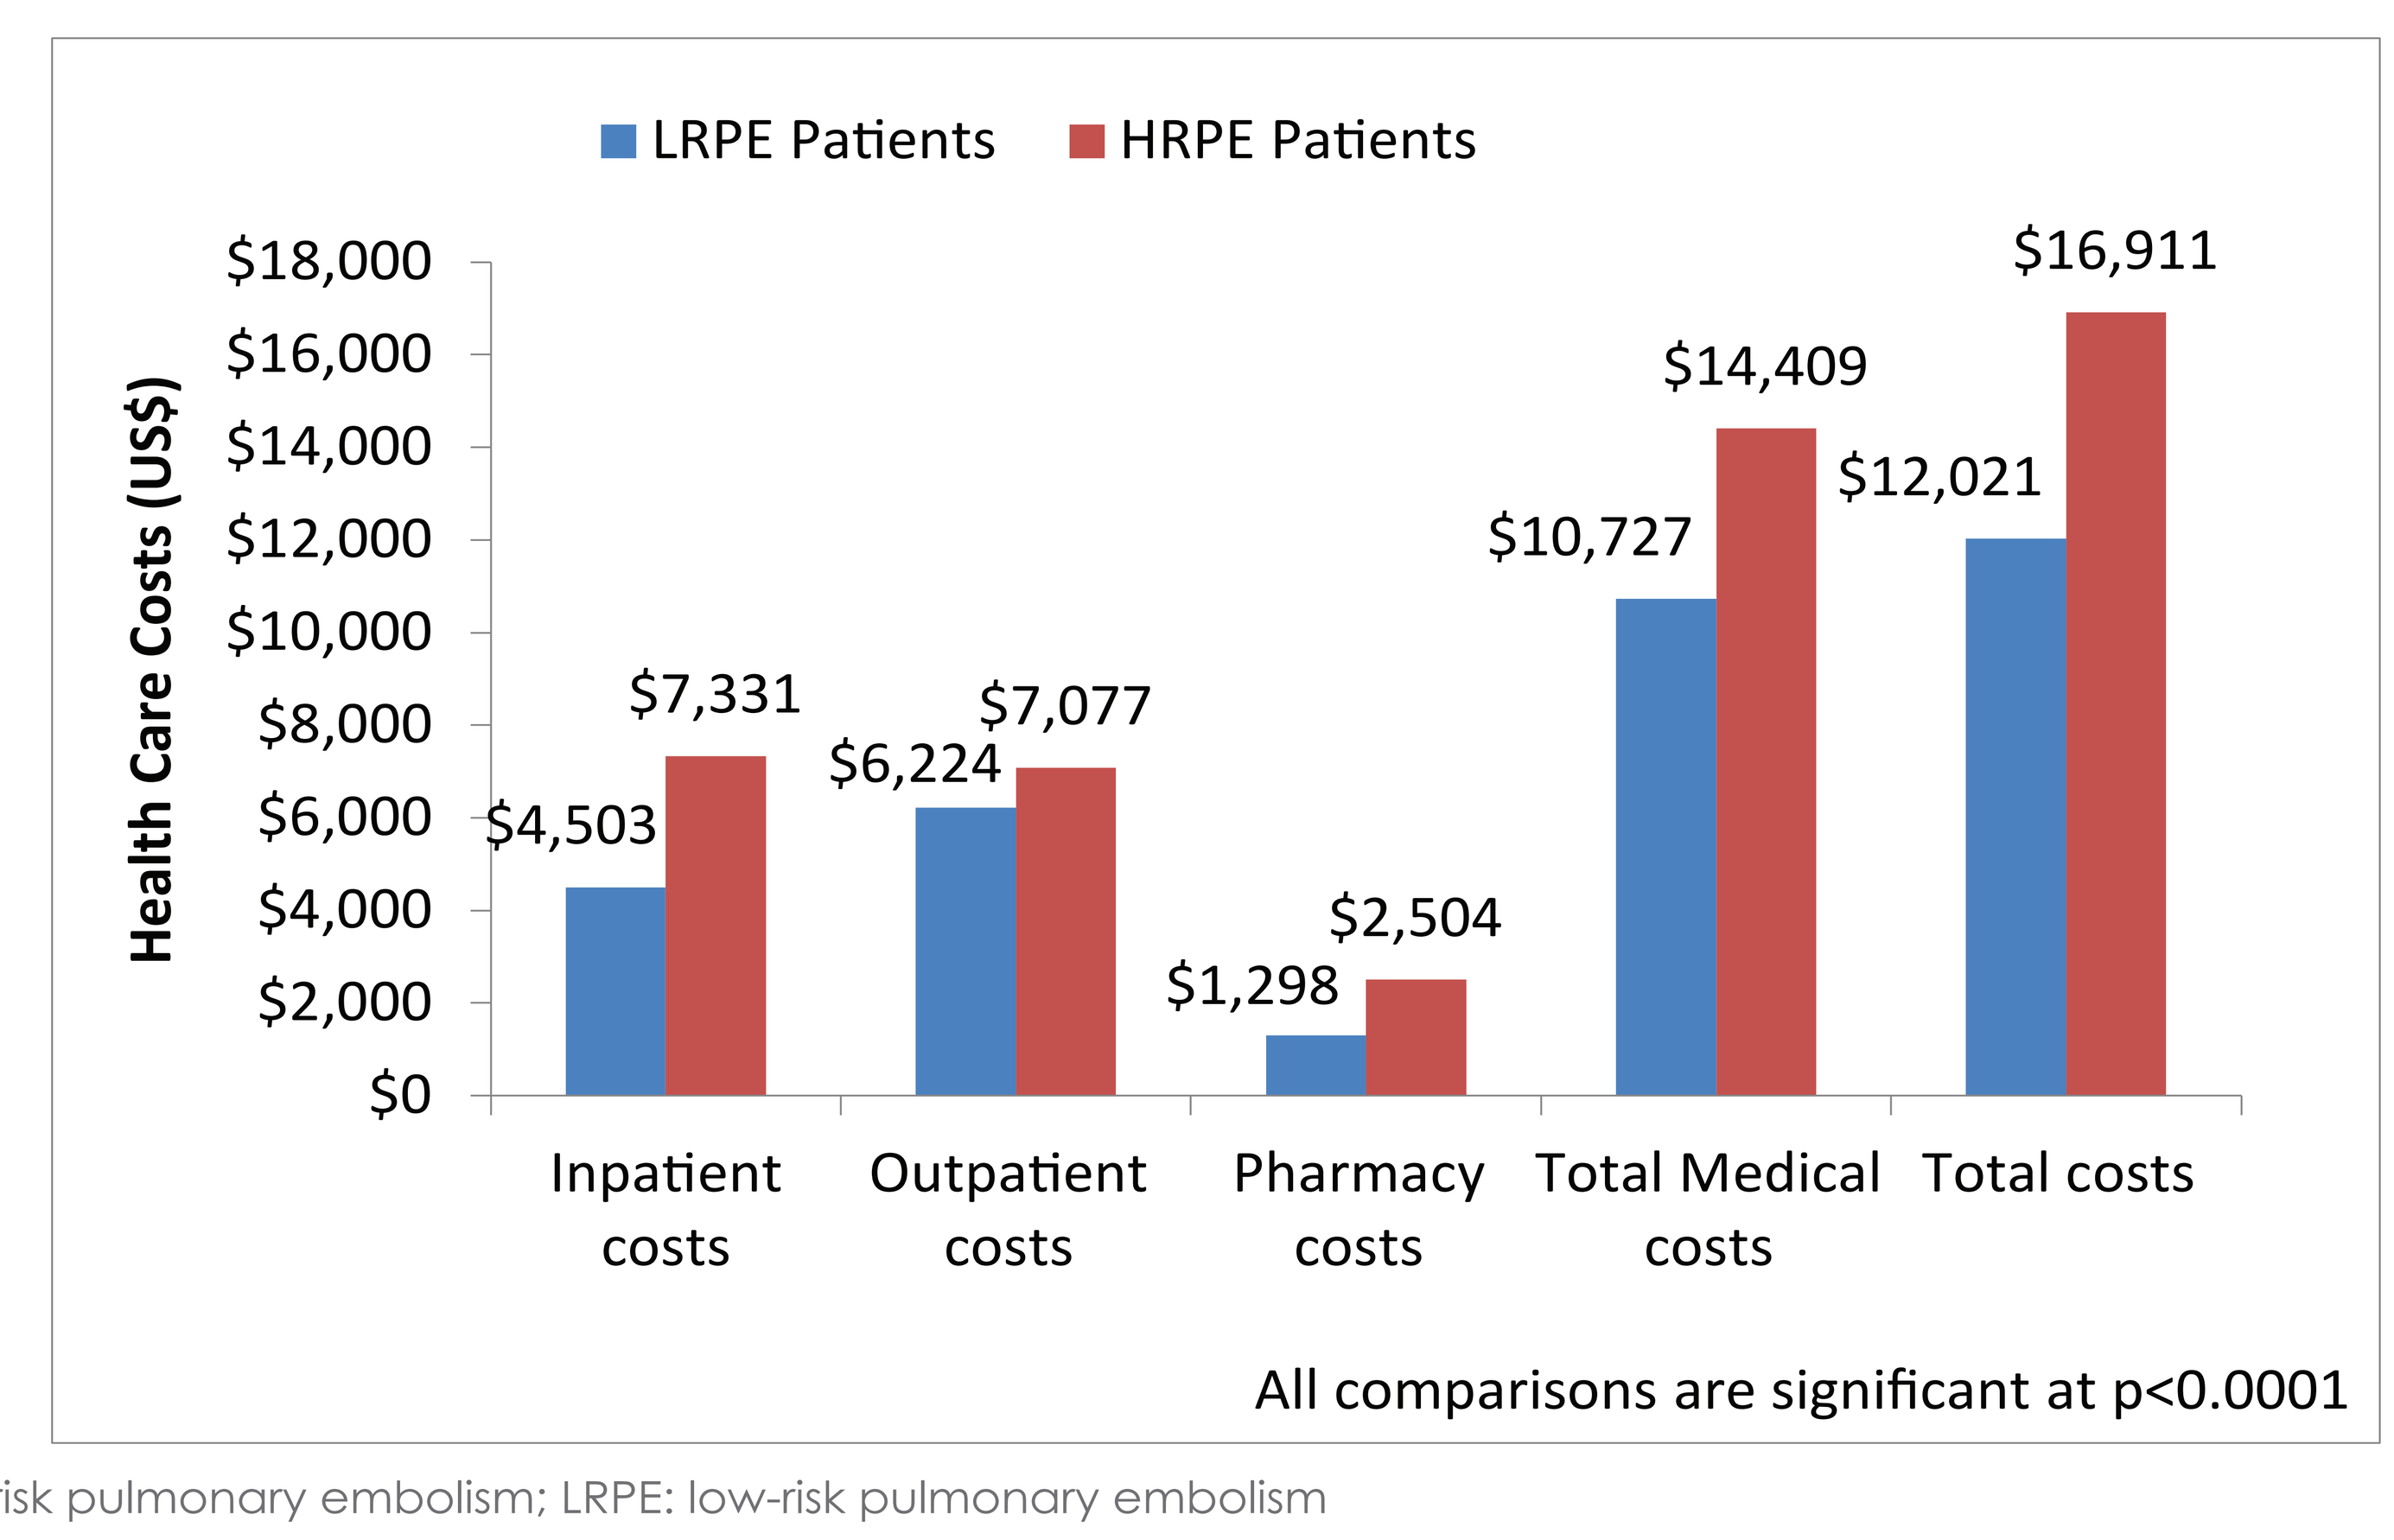
LRPE Patients
HRPE Patients
Health Care Costs (US$)
$18,000
$16,000
$14,000
$12,000
$10,000
$8,000
$6,000
$4,000
$2,000
$0
$4,503
$7,331
$6,224
$7,077
$1,298
$2,504
$10,727
$14,409
$12,021
$16,911
Inpatient costs
Outpatient costs
Pharmacy costs
Total Medical costs
Total costs
All comparisons are significant at p<0.0001

HRPE: high-risk pulmonary embolism; LRPE: low-risk pulmonary embolism

LIMITATIONS

- Inherent limitations of source administrative claims data exist.
- Anticoagulant claims during hospitalization were required to remove rule-out PE diagnoses.
- Only US data from a specific subpopulation were included; general applicability requires further study.

CONCLUSIONS

Using the sPESI score, LRPE patients had lower clinical and economic burden than HRPE patients.

DISCLOSURES

This research was funded by Janssen Scientific Affairs, LLC (Raritan, NJ).

POSTER PRESENTED AT THE *INTERNATIONAL SOCIETY ON THROMBOSIS AND HEMOSTASIS 2017 CONGRESS*; JULY 8-13, 2017; BERLIN, GERMANY

ISTH 2017
795--PB
Cardiovascular Risk Factors
Phil Wells
DOI: 10.3252/pso.eu.ISTH2017.2017
Poster presented at ISTH2017 on:
Tuesday, July 11